CUSA081 (Reteplase) for the Treatment of Central Venous Access Device Occlusions

Michael Wright[1]; Giovanni Piotti[2]

1 Williamson Medical Center, Franklin, TN, USA; 2 Chiesi Farmaceutici S.p.A. Parma, Italy

Background

Introduction

• Dysfunction of Central Venous Access Devices (CVADs) is a serious complication that causes delay in therapy administration, prolonged hospitalization, patient discomfort and increased costs

• Thrombotic occlusion is the most frequent cause of CVAD dysfunction

• Thrombolytic agents successfully restore the patency of occluded CVADs in up to 80% of cases, and are endorsed by current guidelines as a first-line treatment1

• Currently, the tissue plasminogen activator (tPA) Alteplase (Cathflo)2 is the only thrombolytic agent available on the US market approved by the FDA for the restoration of function of CVADs

Reteplase for treatment of dysfunctional CVADs

• Reteplase is a 3rd generation recombinant tPA,

• Reteplase lacks the fibronectin-like finger domain of alteplase that confers high fibrin binding affinity (Fig. 1)

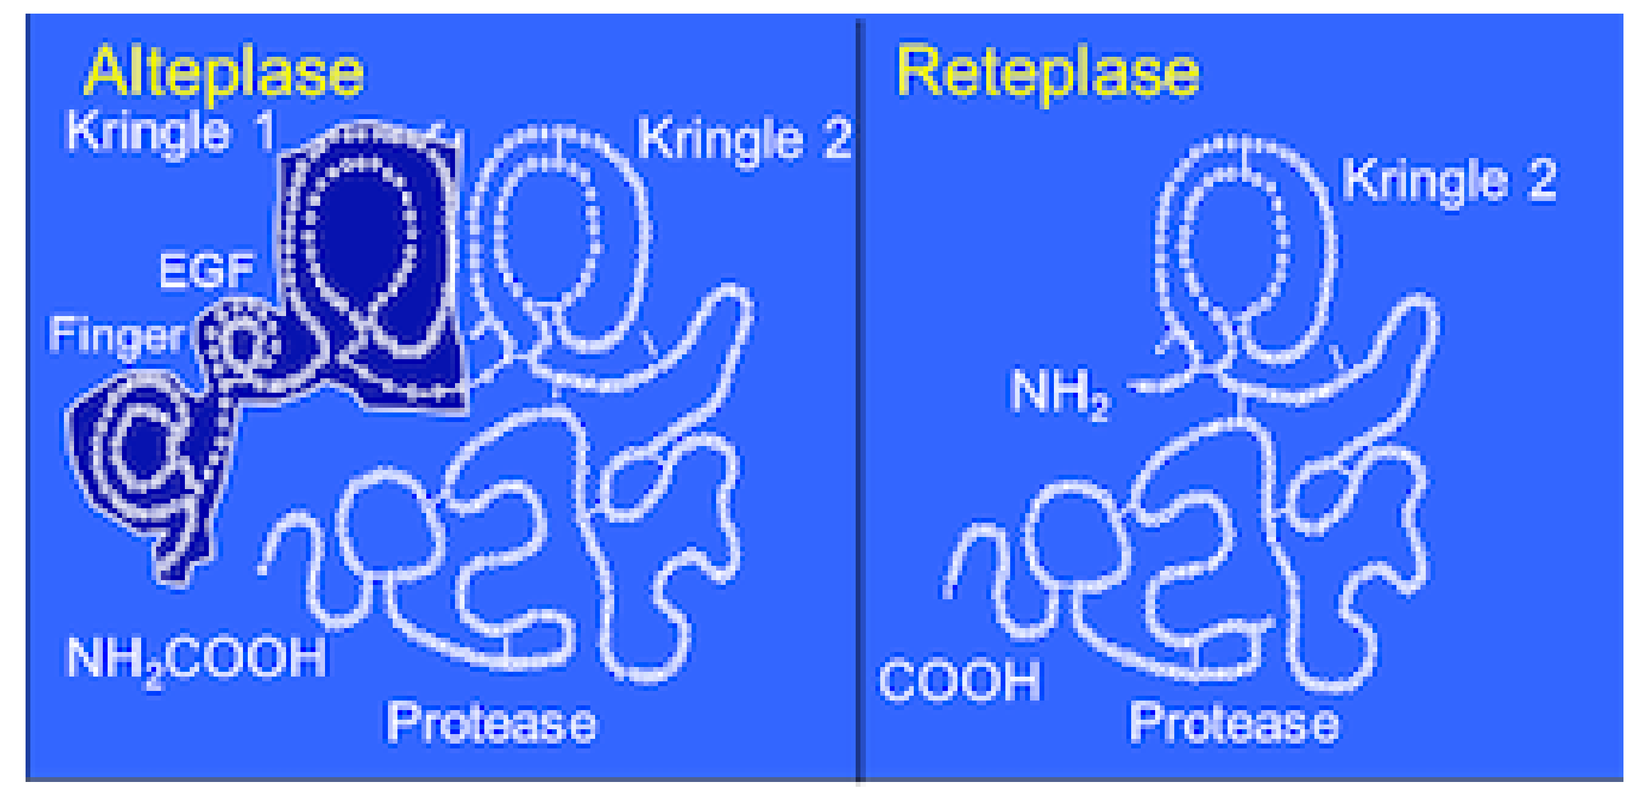
Alteplase
Kringle 1
Kringle 2
EGF
Finger
NH_2COOH
Protease
Reteplase
Kringle 2
NH_2
COOH
Protease

Figure 1. Molecular structure of alteplase compared to reteplase

• Compared to alteplase, reteplase binds less tightly to fibrin, thus penetrating deeper in the clot and potentially being more rapid in lysing clots3

• Preliminary studies found that reteplase is highly effective at restoring CVAD patency when instilled within the catheter at a dose of 0.4 units46

• Chiesi Famaceutici S.p.A. (Chiesi) has defined a clinical development program, namely READY, composed of 2 Phase3 studies and 1 pediatric study with the aim to develop reteplase for the restoration of function to dysfunctional CVADs

• The program has been agreed with the FDA

• The formulation of reteplase under development for this indication is CUSA-081 (0.4 units of reteplase/2 ml)

READY-1: Study Design

• READY 1 is a phase III, multicenter, randomized, double-blind, parallel-group, active- and placebo-controlled study examining the use of CUSA-081 (reteplase) vs placebo or alteplase for the treatment of dysfunctional non-hemodialysis CVADs

• It is sponsored by Chiesi Famaceutici S.p.A. (NCT03594175)

• It plans to randomize 841 adults across 100 sites in 8 countries in Europe, South, and North America (Fig. 2)

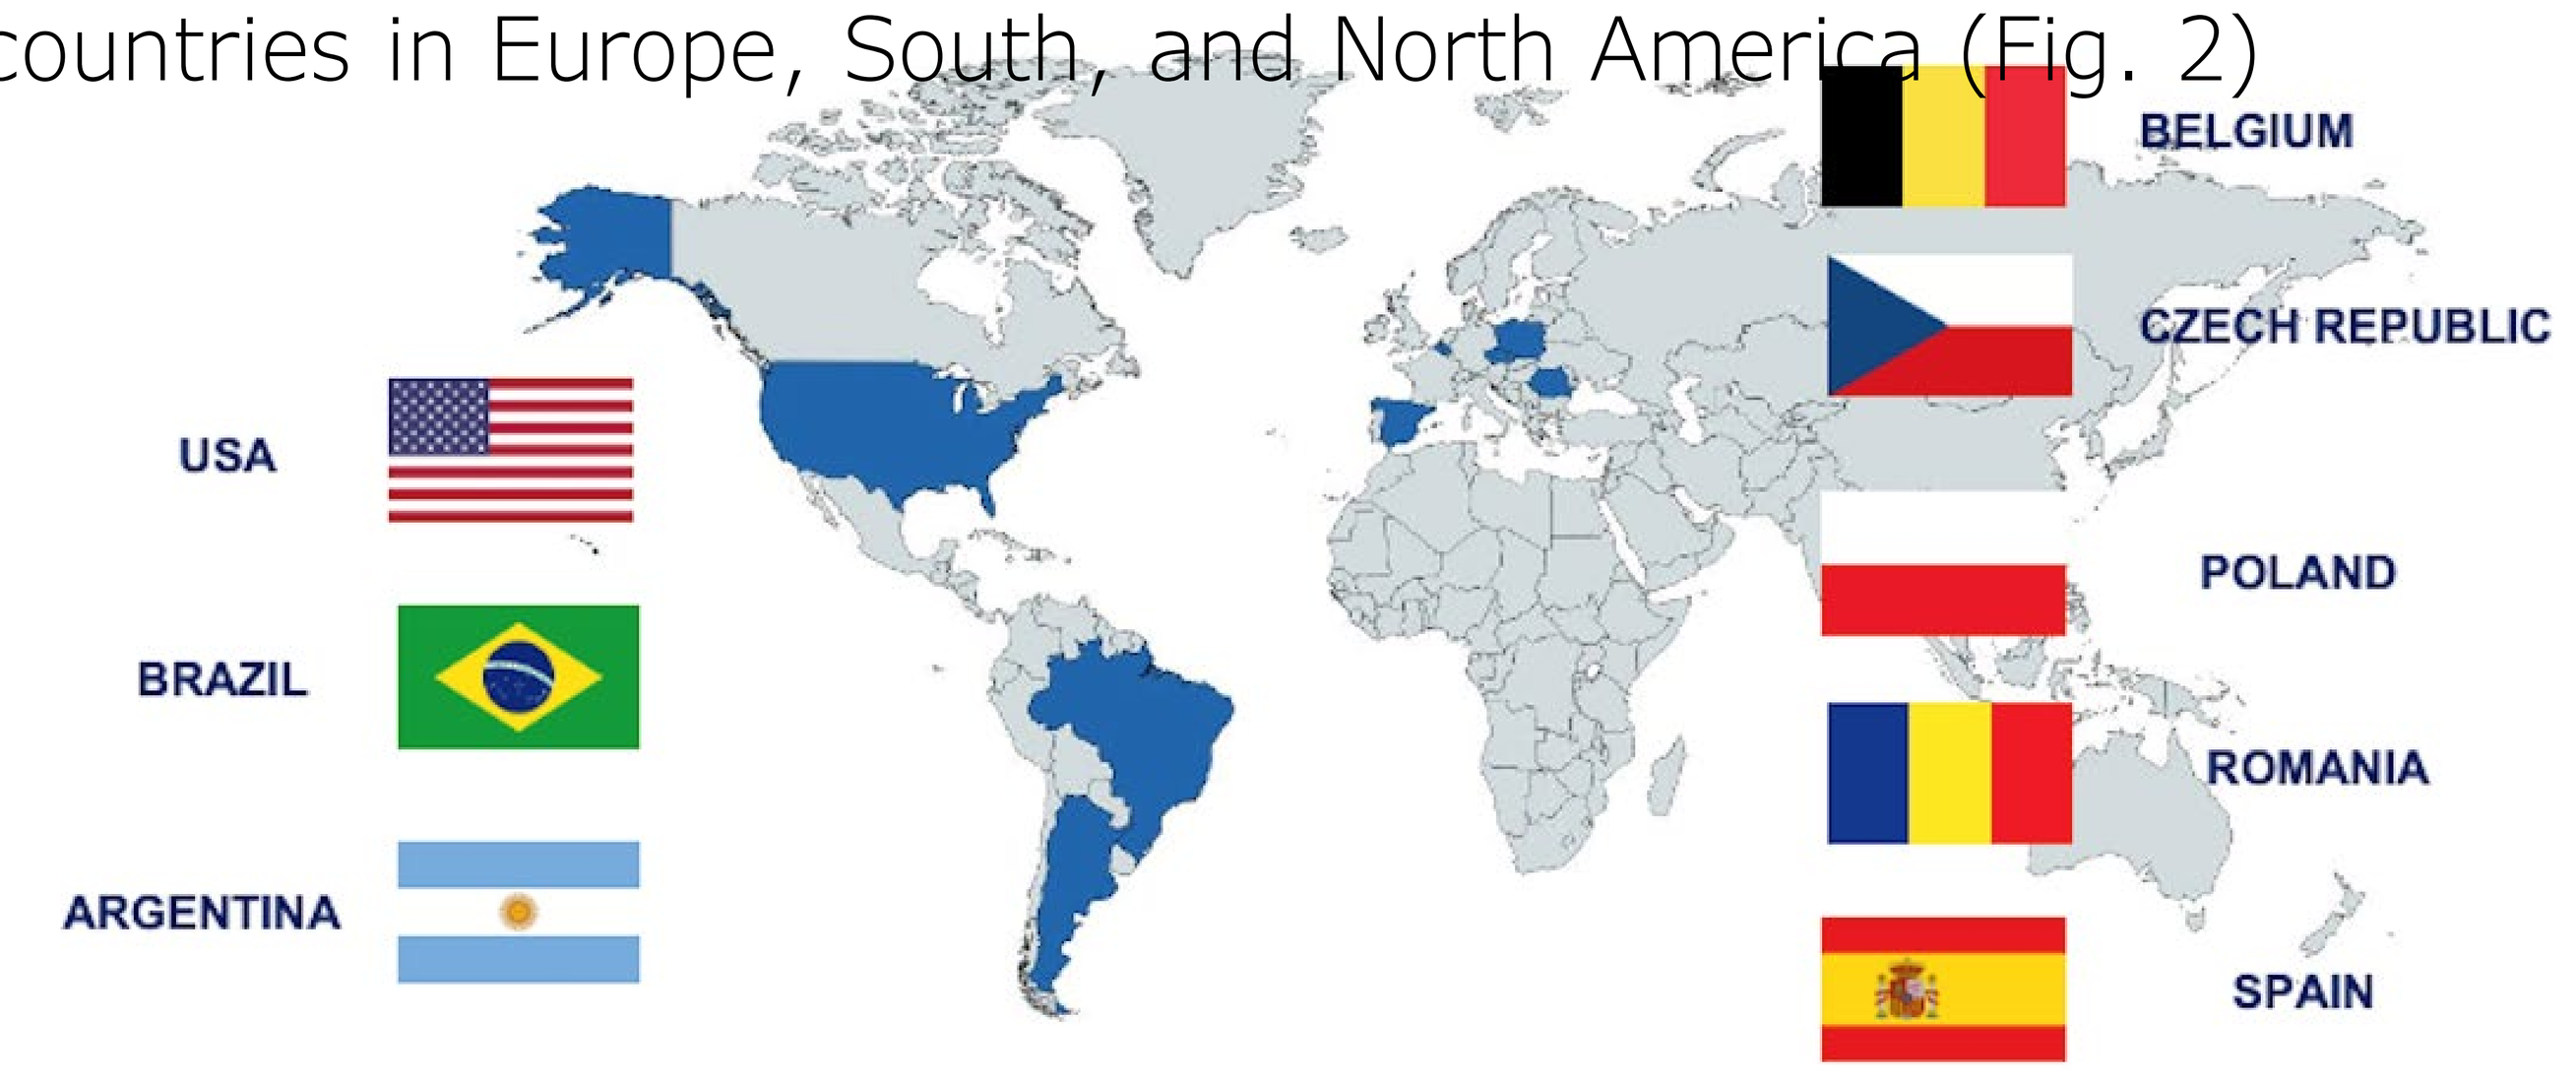
USA
BRAZIL
ARGENTINA
BELGIUM
CZECH REPUBLIC
POLAND
ROMANIA
SPAIN

Figure 2. Study site countries for READY-1 Study

Key inclusion criteria

• Implanted CVAD (single or multi-lumen, implanted port, or peripherally inserted central catheter) in place for > 24 hours and documented as previously patent.

• Inability to have 3 ml of blood withdrawn.

Key exclusion criteria

• CVAD used for hemodialysis.

• CVAD known to be dysfunctional for >48 hours.

• Known non-thrombotic occlusion.

• Known or suspected catheter related blood stream infection.

• Known high risk of bleeding or embolic complication.

Randomization

• Subjects will be randomized after screening and eligibility confirmation with an allocation ratio of 9 to CUSA-081, 1 to placebo, 6 to alteplase (Fig. 3)

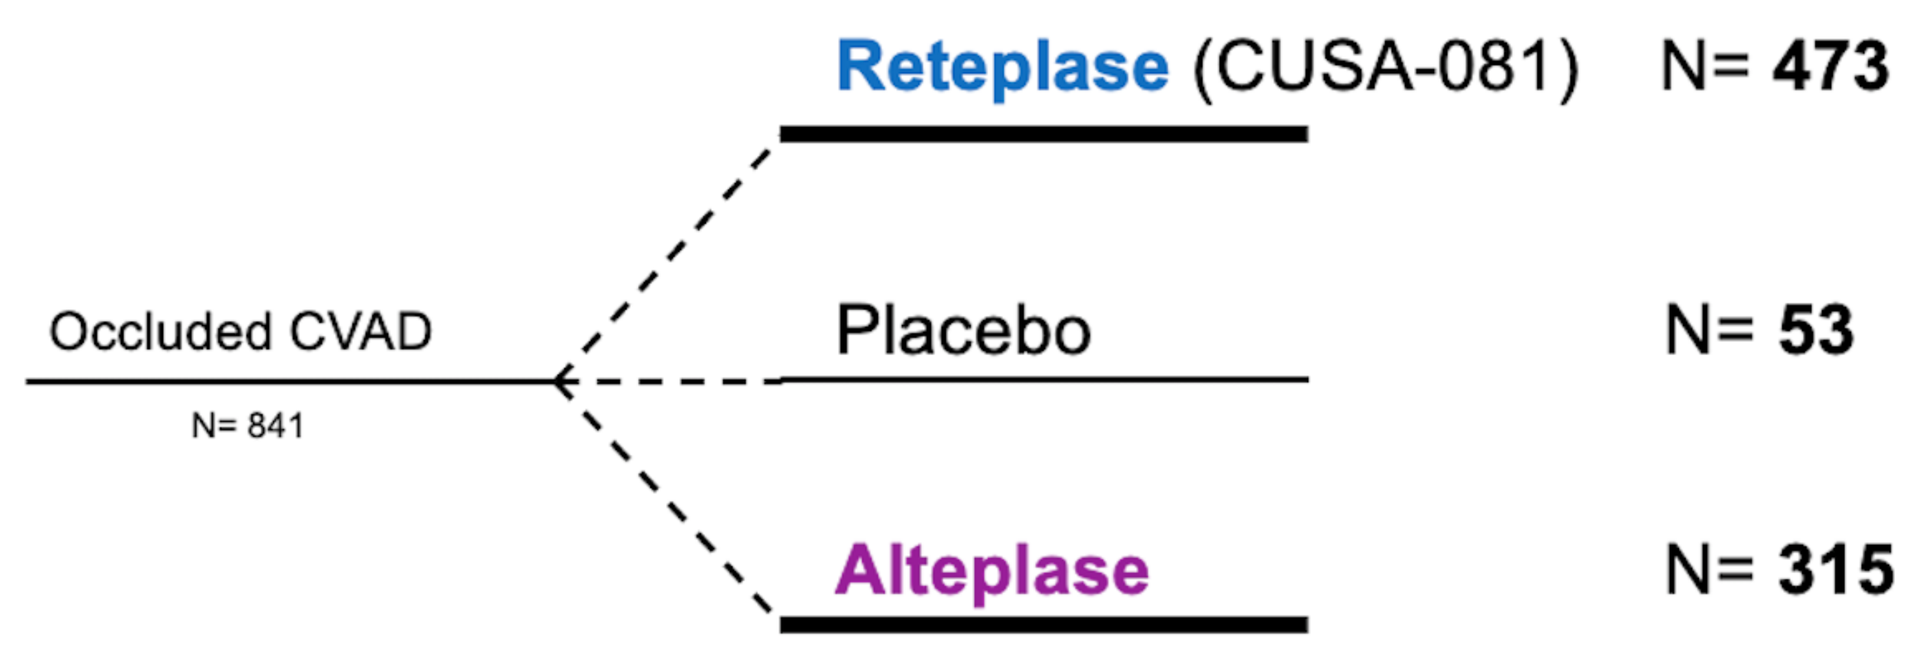
Reteplase (CUSA-081) N= 473
Occluded CVAD
N= 841
Placebo N= 53
Alteplase N= 315

Figure 3. Randomization plan for READY-1

READY-1: Study Methodology

Blinded Study Drug

• Up to 2 intra-catheter instillations of the blinded study drug are foreseen (Fig. 4)

Treatment Arm	Syringe 1 Given at minute 0 Assessed at minutes 30, 60, 90	Syringe 2 Given, if needed, at minute 90 Assessed at minutes 120, 150, 180
A	**CUSA-081** 0.70mg (0.4 units) / 2 mL	**CUSA-081** 0.70mg (0.4 units) / 2 mL
B	**Placebo** 2 mL normal saline (0.9%)	**Placebo** 2 mL normal saline (0.9%)
C	**Alteplase** 2 mg/ 2 mL	**Alteplase** 2 mg/ 2 mL

Figure 4. Investigational drug dosing and sequence

Study Plan and Evaluation of CVAD patency (Fig. 5)

• Restoration of CVAD patency is defined as the ability to have 3 ml of blood withdrawn and 5 ml of saline infused from the dysfunctional CVAD lumen

• Procedures and timepoints for CVAD assessments are indicated in Fig. 5

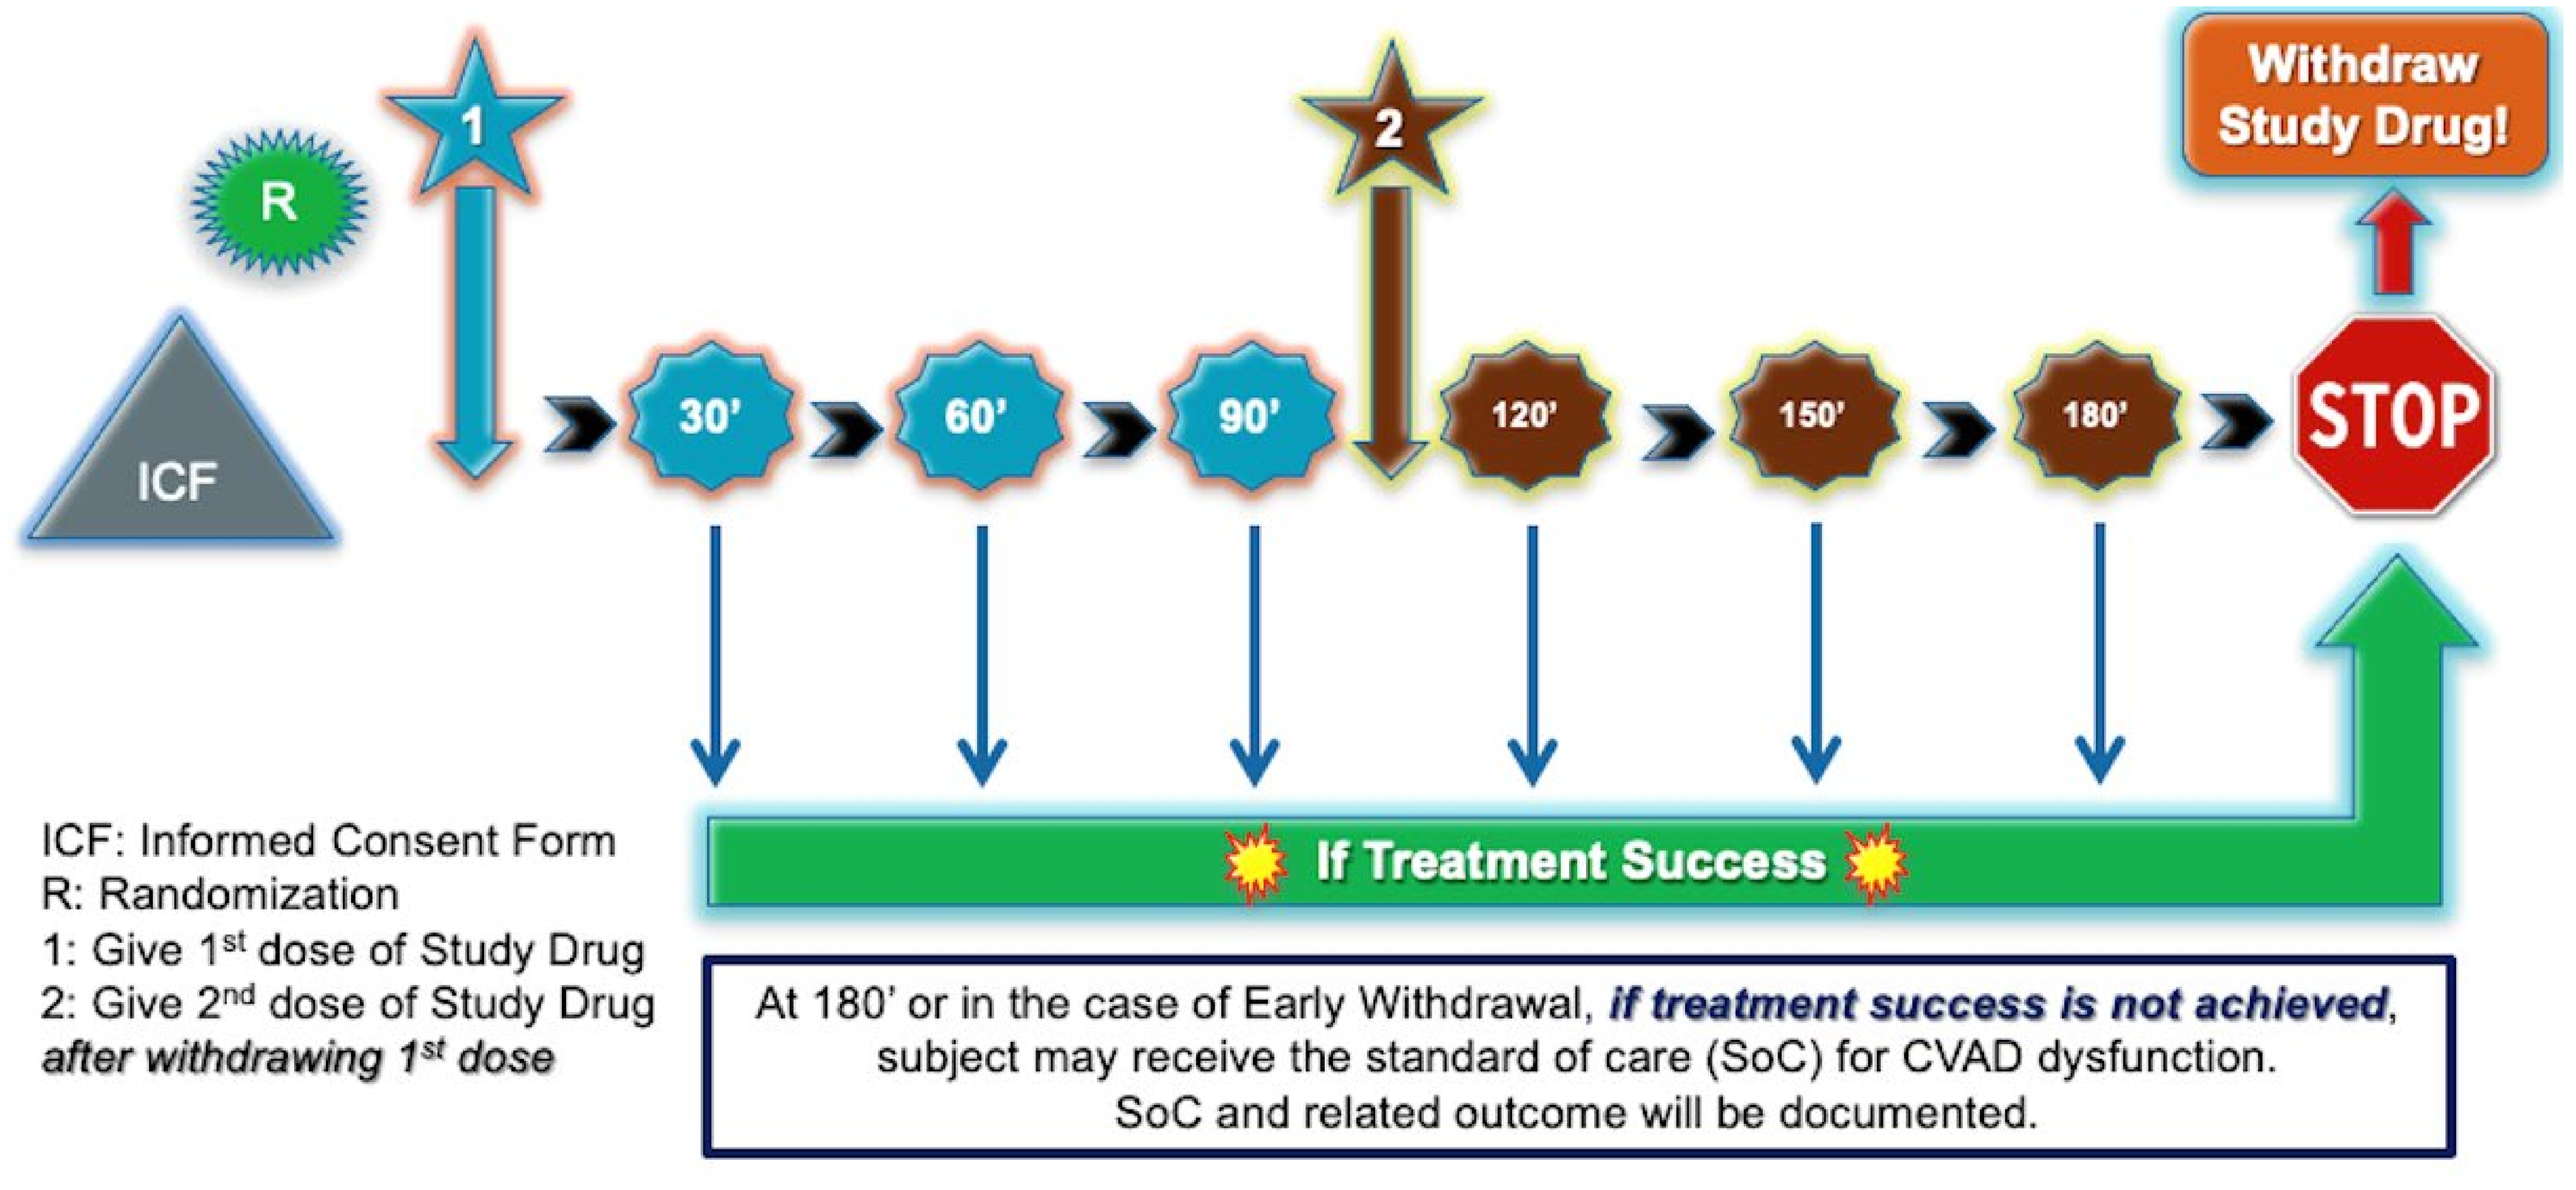
R
1
2
Withdraw Study Drug!
ICF
30'
60'
90'
120'
150'
180'
STOP
ICF: Informed Consent Form
R: Randomization
1: Give 1st dose of Study Drug
2: Give 2nd dose of Study Drug after withdrawing 1st dose
If Treatment Success
At 180' or in the case of Early Withdrawal, if treatment success is not achieved, subject may receive the standard of care (SoC) for CVAD dysfunction.
SoC and related outcome will be documented.

Figure 5. Study Plan and Evaluation of CVAD Patency for READY-1 Study

Primary efficacy endpoint

• To demonstrate the superiority of CUSA-081 compared to placebo in the rate of treatment success following a single administration with a dwell time up to 90 min

Key secondary endpoint

• To demonstrate noninferiority of CUSA-081 vs alteplase with a dwell time up to 90 min

Further important endpoints

• To evaluate the safety and tolerability of CUSA081.

• To evaluate the rate of catheter re-occlusion within 30 days

READY-1: Recruitment Status

• The first patient was randomized in Feb 2020

• Study recruitment has been hindered by COVID pandemic

• 379 participants have been randomized as of 19.Sep.2022

• Study completion is now expected in the first half of 2025 (Fig. 6)

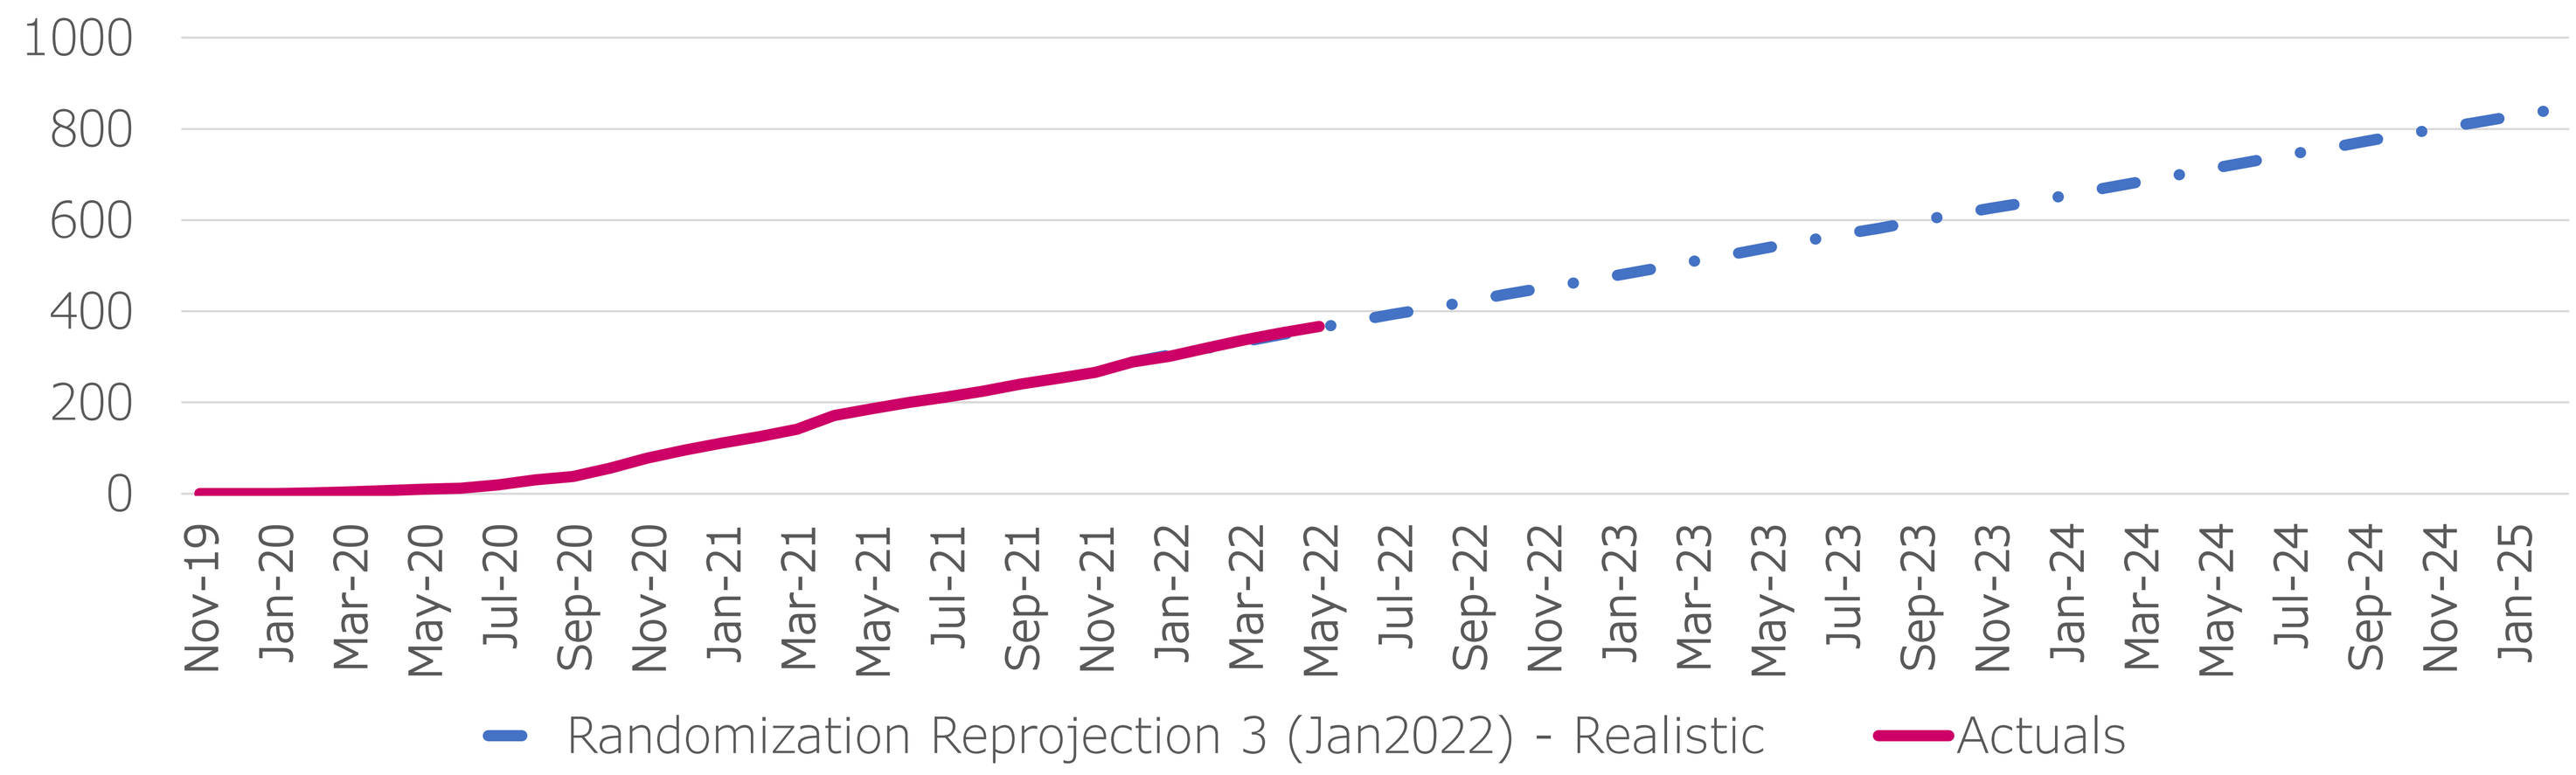
1000
800
600
400
200
0
Nov-19
Jan-20
Mar-20
May-20
Jul-20
Sep-20
Nov-20
Jan-21
Mar-21
May-21
Jul-21
Sep-21
Nov-21
Jan-22
Mar-22
May-22
Jul-22
Sep-22
Nov-22
Jan-23
Mar-23
May-23
Jul-23
Sep-23
Nov-23
Jan-24
Mar-24
May-24
Jul-24
Sep-24
Nov-24
Jan-25
Randomization Reprojection 3 (Jan2022) - Realistic
Actuals

Figure 6. Global READY-1 randomization status

• 26.2% of READY-1 study participants have been randomized in US, 15% in Latin America and in 58.8% in Europe (Fig. 7)

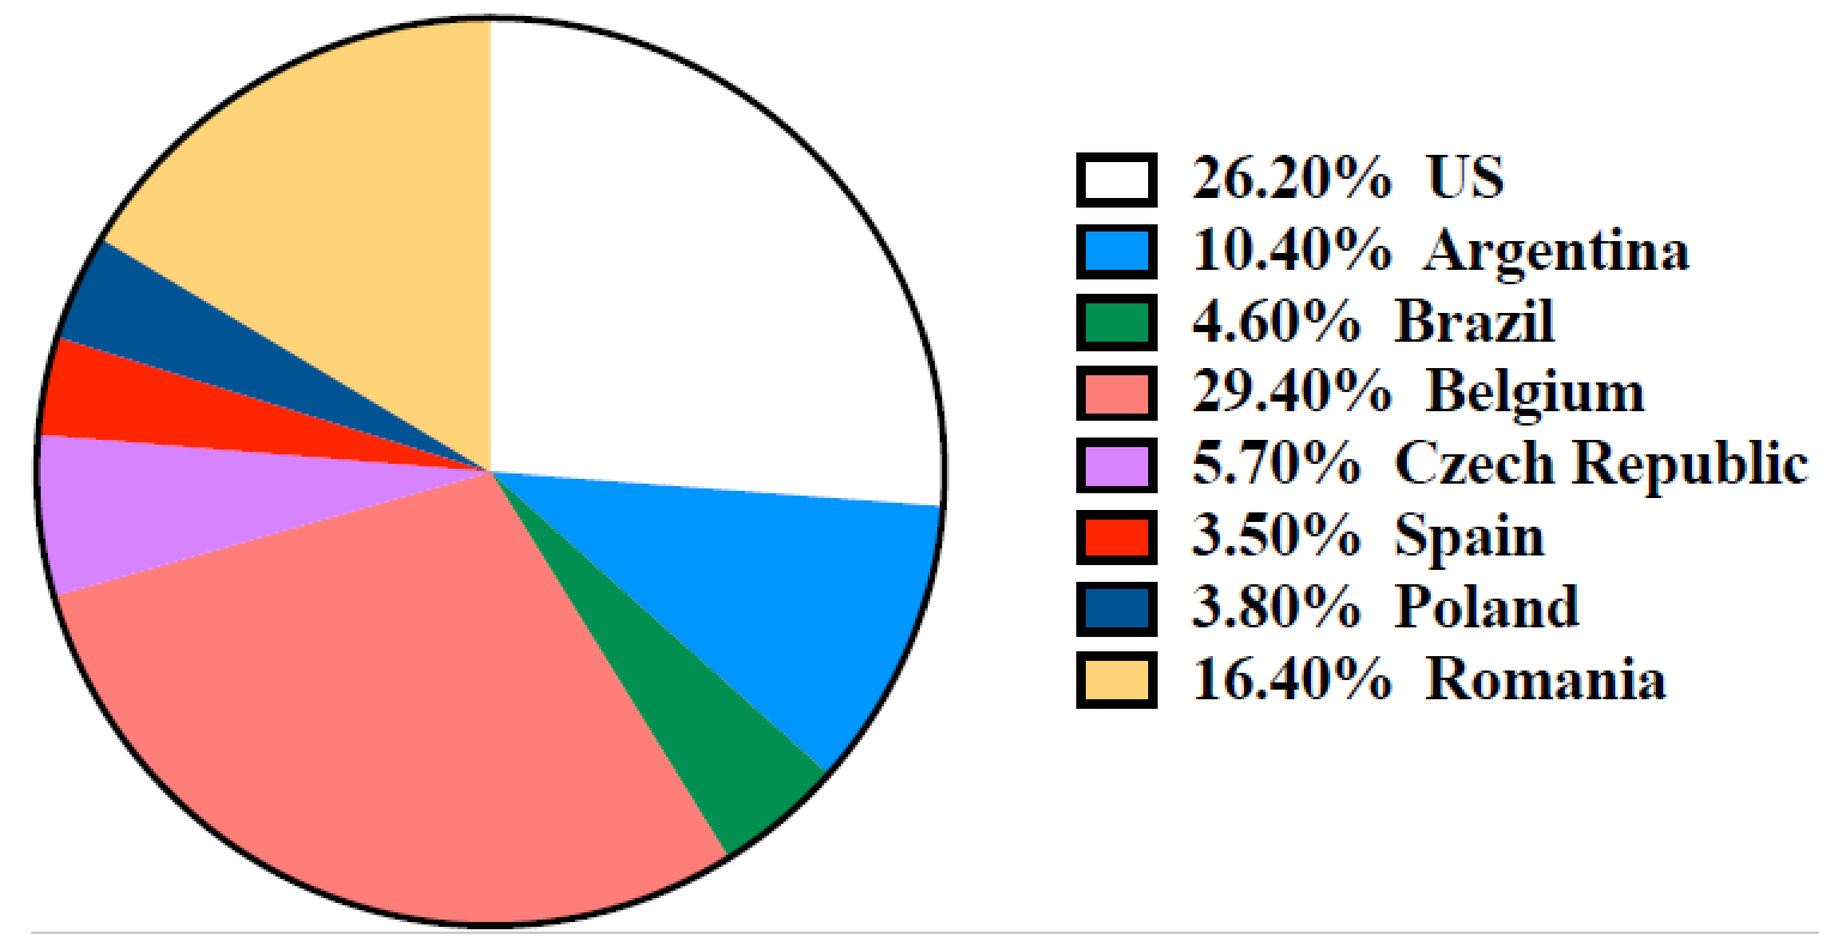
26.20% US
10.40% Argentina
4.60% Brazil
29.40% Belgium
5.70% Czech Republic
3.50% Spain
3.80% Poland
16.40% Romania

Figure 7. Proportion of READY-1 participants by Country

References

1. Ernst FR, Chen E, Lipkin C, et al. Comparison of Hospital Length of Stay, Costs, and Readmission of Alteplase Versus Catheter Replacement Among Patients With Occluded Central Venous Catheters. J Hosp Med. 2014;9(8):4906.
2. Baskin JL, Pui C, Reiss U, et al. Management of occlusion and thrombosis associated with longterm indwelling central venous catheters. Lancet. 2009;374(9684):159169.
3. Smalling RW. Molecular Biology of Plasminogen Activators: What are the Clinical Implications of Drug Design? Am J Cardiol. 1996;78(suppl 12A):27.
4. Owens L. Reteplase for clearance of occluded venous catheters. Am J HealthSyst Pharm. 2002; 59(17):163840.
5. Liu CY, Jain V, Shields AF, et al. Efficacy and Safety of Reteplase for Central Venous Catheter Occlusion in Patients with Cancer. J Vasc Interv Radiol. 2004;15:3944.
6. DaCosta AC, Ribiero JM, Vasques CI, et al. Interventions to obstructive longterm central venous catheter in cancer patients: a meta-analysis. Support Care Cancer. 2019;27:407421.