MM - 1223

Arlocabtagene autoleucel (arlo-cel), a GPRC5D-targeted CAR T cell therapy for patients with relapsed/refractory multiple myeloma: updated phase 1 safety and efficacy results in patients with 1-3 prior regimens

Susan Bal,[1] Myo Htut,[2] Jesus G. Berdeja,[3] Larry D. Anderson Jr,[4] Adriana C. Rossi,[5] Tara Gregory,[6] Luciano J. Costa,[1] Hongxiang Hu,[7] Ziyan Guo,[7] Jinjie Chen,[7] Kristina M. Jordahl,[7] Naomey Sarkis,[7] Safiyyah Ziyad,[7] Wei-Ming Kao,[7] Allison J. Kaeding,[7]* Michael R. Burgess,[7] Omar Nadeem[8]

[1]University of Alabama at Birmingham, Birmingham, AL, USA; [2]City of Hope Comprehensive Cancer Center, Duarte, CA, USA; [3]Greco-Hainsworth Tennessee Oncology Centers for Research, Nashville, TN, USA; [4]Hematologic Malignancies and Cellular Therapy Program, Simmons Comprehensive Cancer Center, UT Southwestern Medical Center, Dallas, TX, USA; [5]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [6]Colorado Blood Cancer Institute, Sarah Cannon Cancer Network, Denver, CO, USA; [7]Bristol Myers Squibb, Princeton, NJ, USA; [8]Dana-Farber Cancer Institute, Boston, MA, USA
*At time of study.

Study summary: phase 1 study (NCT04674813) — cohort C

How was the study designed?

Cohort C of this phase 1 study tested a dose of 150 million cells of arlo-cel given to adults who previously received 1-3 lines of therapy for their relapsed/refractory multiple myeloma (RRMM)

Who took part in this study?

31 people who previously received 1-3 lines of therapy for their RRMM joined the study and had their white blood cells collected; all 31 received arlo-cel

2 was the median number of previous treatments people had tried for their MM

>54% of people did not respond to at least 3 different types of myeloma drugs taken before the study began

What were the key results of the study?

Safety: side effects were similar to previous reports of arlo-cel

- No people had severe or disabling side effects such as infections or cytokine release syndrome
- Side effects in the skin, nails, and mouth related to arlo-cel were short-lived; most resolved with minimal treatment
- Some people experienced effects on their brain or nervous system

Efficacy: arlo-cel showed strong and long-lasting benefits

- **Overall response rate:** 94% of people responded to arlo-cel
- **Progression-free survival:** 63% of people had no worsening myeloma within 18 months of receiving arlo-cel
- **Overall survival:** 100% of people were alive 18 months after receiving arlo-cel

What ongoing trials are currently studying arlo-cel?

- QUINTESSENTIAL (NCT06297226) — **Phase 2 study:** arlo-cel is being studied in people who previously received at least 4 types of treatments and had at least 3 lines of therapy
- QUINTESSENTIAL-2 (NCT06615479) — **Phase 3 study:** arlo-cel is being compared with standard treatment in people who had previously received 1-3 lines of therapy, including lenalidomide

Patients demonstrated deep, durable responses following a single infusion of arlo-cel

Figure 3. Response to arlo-cel infusion

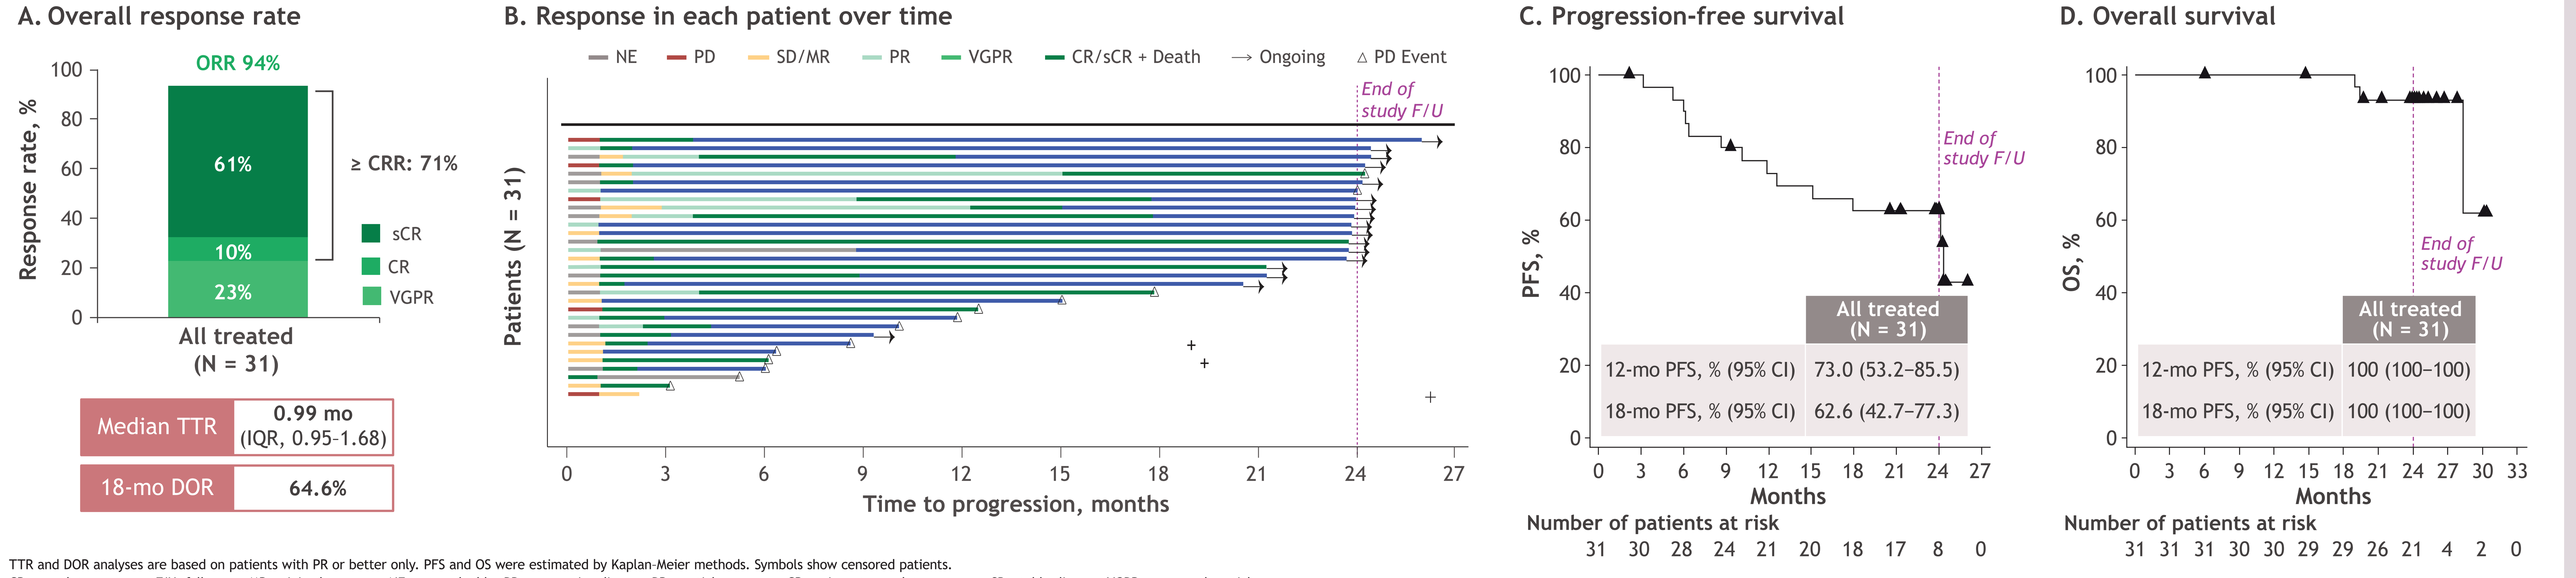
A. Overall response rate
ORR 94%
61%
10%
23%
≥ CRR: 71%
sCR
CR
VGPR
Response rate, %
All treated (N = 31)
Median TTR 0.99 mo (IQR, 0.95-1.68)
18-mo DOR 64.6%
B. Response in each patient over time
NE
PD
SD/MR
PR
VGPR
CR/sCR + Death
Ongoing
PD Event
End of study F/U
Patients (N = 31)
Time to progression, months
C. Progression-free survival
PFS, %
End of study F/U
All treated (N = 31)
12-mo PFS, % (95% CI) 73.0 (53.2–85.5)
18-mo PFS, % (95% CI) 62.6 (42.7–77.3)
Months
Number of patients at risk
31 30 28 24 21 20 18 17 8 0
D. Overall survival
OS, %
End of study F/U
All treated (N = 31)
12-mo PFS, % (95% CI) 100 (100–100)
18-mo PFS, % (95% CI) 100 (100–100)
Months
Number of patients at risk
31 31 31 30 30 29 29 26 21 4 2 0

TTR and DOR analyses are based on patients with PR or better only. PFS and OS were estimated by Kaplan-Meier methods. Symbols show censored patients.
CR, complete response; F/U, follow-up; MR, minimal response; NE, not evaluable; PD, progressive disease; PR, partial response; sCR, stringent complete response; SD, stable disease; VGPR, very good partial response.

Background

- Multiple myeloma is a hematologic malignancy characterized by frequent relapses in patients with shorter responses and worsening prognosis with each regimen, even within early lines of therapy[1-4]
- B-cell maturation antigen (BCMA)-targeting agents are becoming increasingly common standard treatments in early-line RRMM
- However, gaps remain owing to target antigen loss, T-cell exhaustion, and significant infection rates, underscoring a high unmet need[1-3]
- G protein-coupled receptor class C group 5 member D (GPRC5D), a validated therapeutic target in MM, exhibits a restricted expression profile with high expression on malignant plasma cells[5,6]
- Arlocabtagene autoleucel (arlo-cel) is an investigational chimeric antigen receptor (CAR) T cell therapy targeting GPRC5D that has demonstrated deep and durable responses with a tolerable safety profile across early- and late-line patients with RRMM[7,8]
- Here we report the final analysis from cohort C of the first-in-human phase 1 study in early-line patients (1–3 prior lines of therapy [pLOT]) with RRMM (data cutoff: February 17, 2026)

Methods

- Cohort C included patients with RRMM who had recieved 1–3 prior lines of therapy; safety and efficacy were assessed in the all-treated and efficacy-evaluable populations (**Figure 1**)

Figure 1. Study design

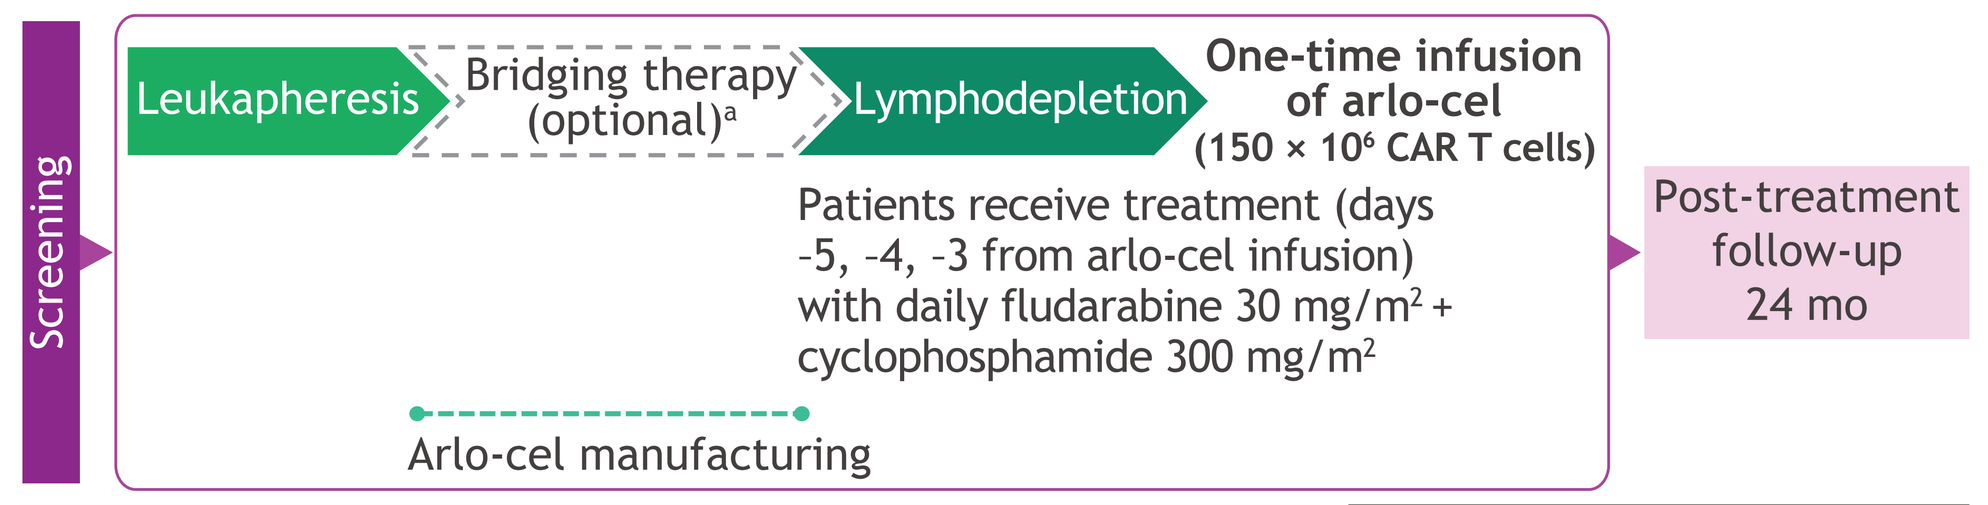
Screening
Leukapheresis
Bridging therapy (optional)a
Lymphodepletion
One-time infusion of arlo-cel (150 × 10^6 CAR T cells)
Patients receive treatment (days -5, -4, -3 from arlo-cel infusion) with daily fludarabine 30 mg/m2 + cyclophosphamide 300 mg/m2
Arlo-cel manufacturing
Post-treatment follow-up 24 mo

Key eligibility criteria - cohort C
- ≥ 18 years of age
- Diagnosis of RRMM with progressive disease ≤ 12 months from completion of most recent regimen
- ECOG performance status 0 or 1
- 1–3 prior anti-myeloma regimens, including a PI and an IMiD
 - Anti-CD38 therapy was not required
 - BCMA-targeted therapies, including CAR T cells, were allowed

Primary objective
- Safety and tolerability

Secondary objectives
- ORR, CRR, DOR, and TTR by IMWG criteria
- PFS
- OS

Exploratory objectives
- MRD-negative status
- Pharmacodynamics

[a]If required for disease control.
All-treated population includes patients who received arlo-cel and efficacy-evaluable population includes patients who received arlo-cel, had measurable disease at last assessment prior to arlo-cel infusion, and had ≥ 1 post infusion disease response assessment.
CRR, complete response rate; DOR, duration of response; ECOG, Eastern Cooperative Oncology Group; IMiD®, immunomodulatory drug; IMWG, International Myeloma Working Group; MRD, minimal residual disease; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PI, proteasome inhibitor; TTR, time to response.

Results

Patients

- The all-treated population comprised 31 patients
 - Arlo-cel manufacturing success rate was 100%; all 31 patients received a dose of 150 × 10^6 cells
- The efficacy-evaluable population (n = 24) excluded 7 patients without measurable disease after bridging therapy
- Median follow-up time was 24.0 (range, 3.8–26.0) months in both the all-treated and efficacy-evaluable populations
- Baseline disease characteristics reflected an early-line population with aggressive disease (**Table 1**)

Table 1. Baseline characteristics

	All treated (N = 31)
Patient characteristics	
Age, median (range), y	62 (31–78)
Sex, male, n (%)	21 (67.7)
Primary race, n (%)	
White	21 (67.7)
Black or African American	6 (19.4)
Other[a]	4 (12.9)
High-risk cytogenetics, n (%)	
del(17p), t(4;14), and/or t(14;16)	8 (25.8)
del(17p)	5 (16.1)
1q21 amp/gain	21 (67.7)
Extramedullary plasmacytoma, n (%)	10 (32.3)
ECOG PS, n (%)	
0	14 (45.2)
1	17 (54.8)
Time since diagnosis, median (range), mo	46.1 (6.2–131.9)
Treatment history	
Any bridging therapy, n (%)	19 (61.3)
Number of prior lines of therapy, median (range)	2 (1–3)
3 prior lines of therapy, n (%)	9 (29.0)
Prior anti-myeloma therapies, n (%)	
Prior IMiD therapy	31 (100)
Prior PI therapy	31 (100)
Prior anti-CD38 therapy	22 (71.0)
Prior BCMA-targeted therapy[b]	1 (3.2)
Refractory status to prior therapy classes,[c] n (%)	
Double-class	27 (87.1)
Triple-class[d]	17 (54.8)
Penta-drug[e]	2 (6.5)
Lenalidomide	28 (90.3)
Refractory to most recent regimen	28 (90.3)

[a]Includes Asian, American Indian or Alaska Native, and unknown racial groups. [b]Belantamab mafodotin. [c]Refractory definition: progression during or within 60 days of end of treatment or lack of response. [d]A PI, IMiD agent, and anti-CD38 antibody. [e]≥ 2 PIs, ≥ 2 IMiD therapies, and an anti-CD38 antibody.

Safety

Treatment-emergent adverse events

- All 31 patients (100%) had ≥ 1 treatment-emergent adverse event (TEAE; **Table 2**)
- Hematologic TEAEs were the most common class of event
 - Prolonged cytopenia (defined as grade 3/4 and not resolving within 1 month of infusion) was reported in 58.1% (18/31) patients; median duration was 63 (range, 50–157) days
- Nonhematologic TEAEs were generally low grade, and there were no grade ≥ 3 infections and infestations

Table 2. Treatment-emergent adverse events

	All treated (N = 31)	
TEAE, n (%)	Any grade	Grade 3/4
Any TEAE[a]	31 (100)	27 (87.1)
Hematologic TEAEs[b]		
Neutropenia	26 (83.9)	25 (80.6)
Thrombocytopenia	22 (71.0)	9 (29.0)
Anemia	14 (45.2)	8 (25.8)
Nonhematologic TEAEs[b]		
Hypocalcemia	13 (41.9)	0
Hyperglycemia	12 (38.7)	1 (3.2)
Dysgeusia[c]	11 (35.5)	—
Hypophosphatemia	10 (32.3)	1 (3.2)
Constipation	10 (32.3)	NR
Infections and infestations[d]	19 (61.3)	0

Medical Dictionary for Regulatory Activities version 28.1; Common Terminology Criteria for Adverse Events version 5.0.
[a]No grade 5 adverse events were reported. [b]≥ 30% in all-treated population for any grade TEAEs. [c]Highest grade possible is grade 2. [d]System organ class; most common preferred terms were upper respiratory infection (25.8%), COVID-19 (9.7%), and pneumonia (9.7%).
NR, not reported.

Adverse events of special interest

- Treatment-related adverse events (TRAEs) occurred in 96.8% (30/31) of patients (**Table 3**)
- All TRAEs of special interest were grade ≤ 2 (**Table 3**)
- Cytokine release syndrome (CRS) was the most common, with median (range) time to onset of 3 (1-4) days and duration of 3 (1-7) days; all events were grade ≤ 2 and resolved
- Immune effector cell–associated neurotoxicity syndrome (ICANS) occurred in 3 patients, with median (range) time to onset of 5 (4-6) days and duration of 2 (1-2) days; all events were grade ≤ 2 and resolved
- Other select neurotoxicity (OSN) occurred in 2 patients; ataxia, neurotoxicity, nystagmus, dysarthria, diplopia, dysmetria, and gait disturbance events qualified as an OSN
- On-target, off-tumor toxicities were primarily grade 1; most resolved without intervention (**Table 3**, **Figure 2**)

Table 3. TRAEs of special interest

	All treated (N = 31)	
TRAE, n (%)	Any grade	Grade 3/4
Any TRAE	30 (96.8)	15 (48.4)
TRAEs of special interest		
CRS	26 (83.9)	0
ICANS	3 (9.7)	0
HLH	0	0
Other select neurotoxicity[a]	2 (6.5)	0
On-target, off-tumor event (cumulative[b])	17 (54.8)	0

Medical Dictionary for Regulatory Activities version 28.1; Common Terminology Criteria for Adverse Events version 5.0. CRS graded using Lee DW, et al.[11]
[a]Includes ataxia, neurotoxicity, nystagmus, dysarthria, diplopia, dysmetria, and gait disturbance (excluding dizziness). [b]Cumulative events include selected preferred terms from skin and subcutaneous tissue disorders and tongue conditions (within gastrointestinal disorders), as well as ad hoc preferred terms (ageusia, dysphagia, dysgeusia, and taste disorder), and excluded preferred terms include night sweats, dermal cyst, hyperhidrosis, and skin mass.
HLH, hemophagocytic lymphohistiocytosis.

Figure 2. Select on-target, off-tumor events

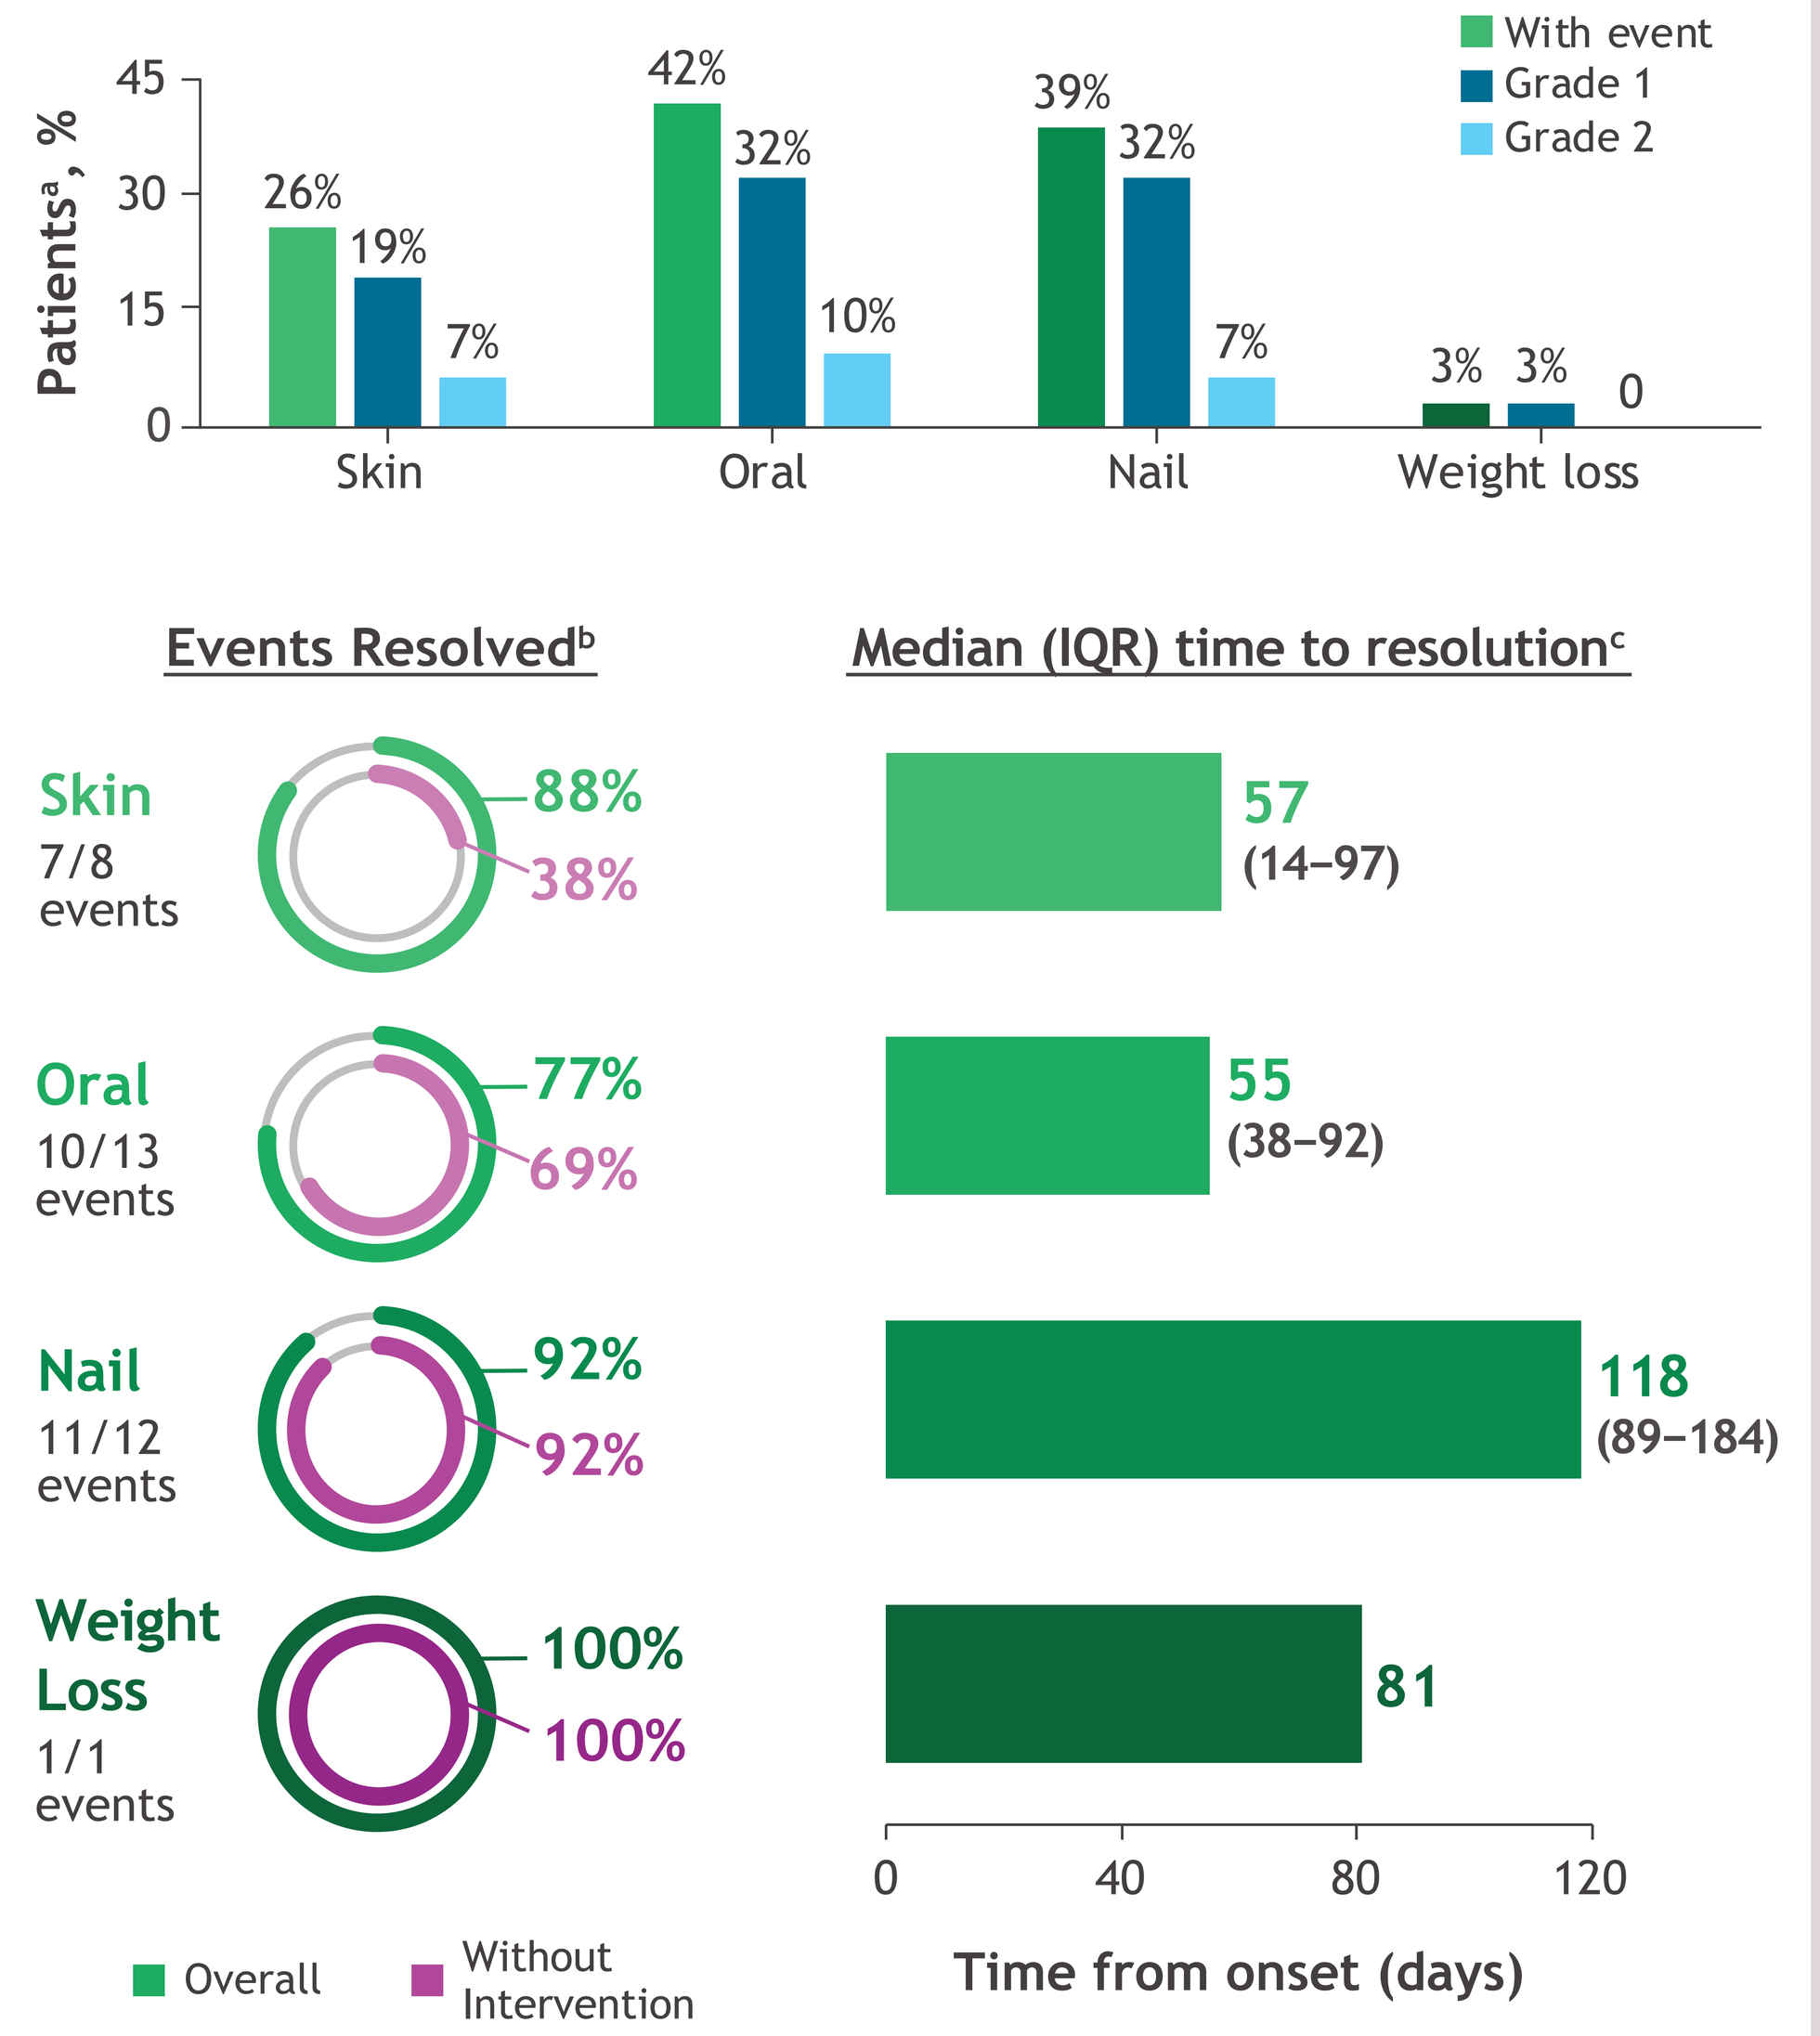
Patients[a], %
26%
19%
7%
42%
32%
10%
39%
32%
7%
3%
3%
0
Skin
Oral
Nail
Weight loss
With event
Grade 1
Grade 2
Events Resolved[b]
Median (IQR) time to resolution[c]
Skin 7/8 events
88%
38%
57 (14–97)
Oral 10/13 events
77%
69%
55 (38–92)
Nail 11/12 events
92%
92%
118 (89–184)
Weight Loss 1/1 events
100%
100%
81
Overall
Without Intervention
Time from onset (days)

Medical Dictionary for Regulatory Activities version 28.1. Common Terminology Criteria for Adverse Events version 5.0.
Multiple events occurring within 1 day from each other are considered as 1 episode.
[a]Percent events calculated out of the all-treated population (N = 31). [b]Percent resolved calculated based on patients with an event. [c]Time to resolution was calculated per episode; only resolved episodes are considered.

Efficacy

- In the all-treated patient population, response rates to arlo-cel were deep and durable (**Figure 3A and B**)
 - ORR was 94% (29/31) and CRR was 71% (22/31)
 - Median TTR was 0.99 (range, 0.95–1.05) months
 - 18-month DOR was 64.6%
- In the efficacy-evaluable population (n = 24), responses were similar
 - ORR was 96% (23/24) and CRR was 67% (16/24)
 - Median TTR was 0.99 (range, 0.95–1.05) months
 - 18-month DOR was 56.5%
- For all treated patients, the 18-month PFS rate was 64.6% and the 18-month OS rate was 100% (**Figure 3C and D**)
- The MRD-negative CRR in MRD-evaluable patients was 65.0% (13/20; 95% CI, 40.8–84.6)

Conclusions

- In this final analysis of cohort C from the phase 1 study of arlo-cel in patients with 1–3 pLOT, the safety profile was consistent with previous reports; no new safety signals were observed
 - No high-grade infections, CRS, ICANS, OSNs, or on-target, off-tumor TEAEs were reported
 - Nonhematologic TEAEs were primarily low grade
- A single infusion of arlo-cel demonstrated deep and durable responses
 - High response rates were observed with an ORR of 94% and CRR of 71%
 - 18-month PFS and OS rates were 62.6% and 100%, respectively
- These results support arlo-cel as a potentially efficacious early-line treatment option for patients with RRMM, with phase 2 (QUINTESSENTIAL [NCT06297226]) and phase 3 (QUINTESSENTIAL-2 [NCT06615479]) trials underway

References

1. Malard F, et al. *Nat Rev Dis Primers*. 2024;10:45. **2.** Goel U, et al. *Blood Cancer J*. 2023;13:11. **3.** Rajkumar SV. *Am J Hematol*. 2024;99(9):1802-1824. **4.** Bal S, et al. *Leukemia*. 2022;36(3):877-880. **5.** Rodriguez-Otero P, et al. *Blood Cancer J*. 2024;14(1):24. **6.** Chari A, et al. *Lancet Haematol*. 2025;12(4):e269-e281. **7.** Bal S, et al. Presented at COMy 2026. Presentation 31. **8.** Bal S, et al. Presented at IMS 2025. Abstract PA-080. **9.** Bal S, et al. Presented at ASH 2023. Abstract 219. **10.** Smith EL, et al. *Sci Transl Med*. 2019;11(485):eaau7746. **11.** Lee DW, et al. *Blood*. 2014;124(2):188-195.

Acknowledgments

- We would like to thank the patients, their families, and the clinical study teams who participated in the trial
- This trial was sponsored by Bristol Myers Squibb
- Medical writing support was provided by Dorothy Dobbins, PhD, CMPP, from Citrus Health Group (Chicago, Illinois), and was funded by Bristol Myers Squibb

Author disclosures

SB: BeOne, Bristol Myers Squibb, Johnson & Johnson - consultancy, research funding, and/or honoraria.

Scientific Content on Demand

To request a copy of this poster:

Scan QR code via a barcode reader application

Presented at the Society of Hematologic Oncology (SOHO) Annual Meeting; September 9–12, 2026; Houston, Texas & Online
Previously presented at the 2026 European Hematology Association (EHA) Congress; June 11–14, 2026; Stockholm, Sweden

Contact: www.globalbmsmedinfo.com

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without written permission from the author of this poster.