SOHO 2026 Annual Meeting | Houston, TX | Poster Presentation | Myeloproliferative Neoplasms

Jefferson Einstein Philadelphia Hospital
Thomas Jefferson University

GLP-1 Receptor Agonists and Major Clinical Outcomes in Adults With Classical Myeloproliferative Neoplasms and Type 2 Diabetes and/or Obesity: A Global Propensity Score-Matched Analysis

Muluken Megiso, MD[1] · Chidiebube Ugwu, MD[1] · Lelo Teka, MD[2] · Dawit Yesho, MD[1] · Tarfa Verinumbe, MD[1] · Angimar Palma, MD[1] · Gabor Varadi, MD[3]

[1] Dept. of Internal Medicine, Jefferson Einstein Philadelphia Hospital, Philadelphia, PA, USA [2] Sheba Medical Center, Tel Aviv, Israel [3] Dept. of Hematology & Oncology, Thomas Jefferson University, Philadelphia, PA, USA

BACKGROUND

Classical MPNs (polycythemia vera, essential thrombocythemia, myelofibrosis) carry substantial cardiovascular and thrombotic morbidity, frequent hospitalization, and premature mortality.

Type 2 diabetes (T2DM) and obesity are common comorbidities in MPN, amplifying thrombotic and cardiovascular risk.

GLP-1 receptor agonists (GLP-1RAs) reduce CV events and mortality in general T2DM/obesity populations (LEADER, SUSTAIN-6 trials).

Anti-inflammatory and anti-thrombotic pleiotropic effects may confer additional benefit in MPN.

No prior large-scale study has evaluated GLP-1RA outcomes specifically in classical MPN.

OBJECTIVE

Evaluate the association of GLP-1RA exposure with major clinical outcomes in adults with classical MPN and T2DM and/or obesity using a global real-world database.

METHODS

DATA SOURCE

TriNetX Global Collaborative Network — federated, de-identified EHR data from academic medical centers worldwide.

STUDY DESIGN

Retrospective cohort study with 1:1 propensity score matching (PSM). Index date = first GLP-1RA prescription (exposed) or first non-GLP-1RA antidiabetic agent (comparator).

ELIGIBILITY & FOLLOW-UP

- Adults 18–90 years with classical MPN (PV, ET, PMF)
- Concurrent T2DM and/or obesity diagnosis
- Outcomes assessed: 30 days to 5 years post-index

PSM VARIABLES

Age, sex, race, BMI, HbA1c, MPN subtype, prior CV disease, anticoagulation use, cytoreductive therapy, and relevant comorbidities.

1,837 GLP-1RA Cohort (post-PSM)	1,837 Comparator Cohort (post-PSM)
541 days Median Follow-up (GLP-1RA)	442 days Median Follow-up (Comparator)

RESULTS — OUTCOME SUMMARY TABLE

Outcome	GLP-1RA (%)	Comparator (%)	HR	95% CI	p-value
All-Cause Mortality	1.0%	4.3%	0.23	0.14–0.39	<0.001
Heart Failure	1.6%	4.3%	0.37	0.24–0.58	<0.001
Ischemic Heart Disease	4.0%	7.7%	0.49	0.36–0.67	<0.001
All-Cause Hospitalization	5.4%	10.4%	0.51	0.39–0.67	<0.001
Venous Thromboembolism	1.9%	3.0%	0.59	0.38–0.92	0.020
Cerebral Infarction	0.6%	1.1%	0.55	0.26–1.15	0.108 (NS)

HR = Hazard Ratio | CI = Confidence Interval | NS = Not Significant | N = 1,837 per cohort after propensity score matching

FOREST PLOT — HAZARD RATIOS (95% CI)

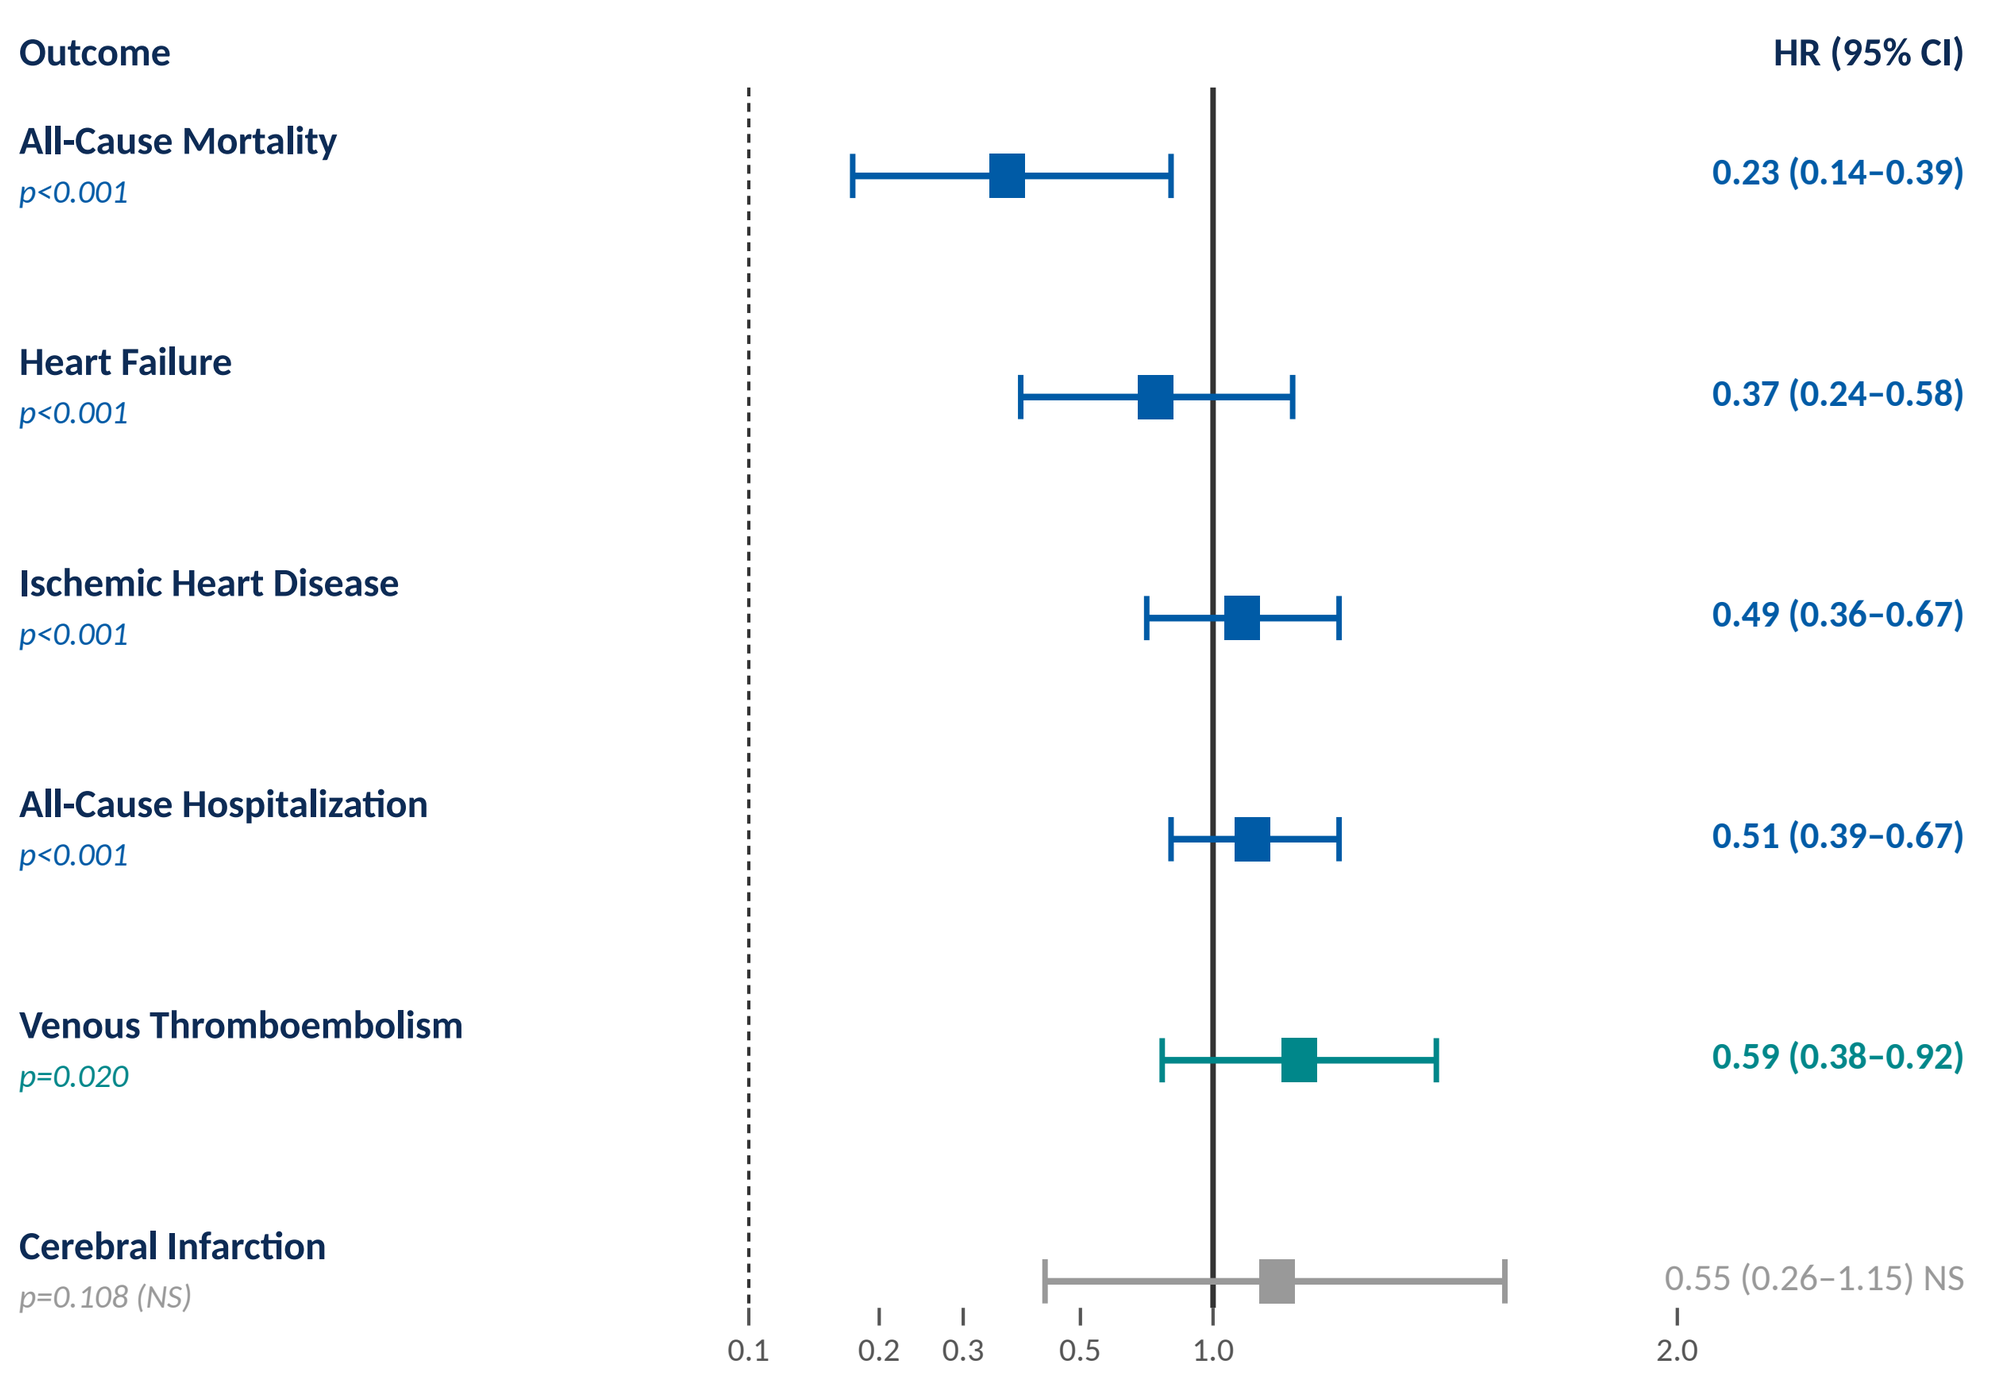
Outcome
HR (95% CI)
All-Cause Mortality
p<0.001
0.23 (0.14–0.39)
Heart Failure
p<0.001
0.37 (0.24–0.58)
Ischemic Heart Disease
p<0.001
0.49 (0.36–0.67)
All-Cause Hospitalization
p<0.001
0.51 (0.39–0.67)
Venous Thromboembolism
p=0.020
0.59 (0.38–0.92)
Cerebral Infarction
p=0.108 (NS)
0.55 (0.26–1.15) NS
0.1 0.2 0.3 0.5 1.0 2.0

HR = Hazard Ratio | CI = Confidence Interval | NS = Not Significant | Null line at HR = 1.0 | N = 1,837 per cohort

EVENT RATE COMPARISON (%)

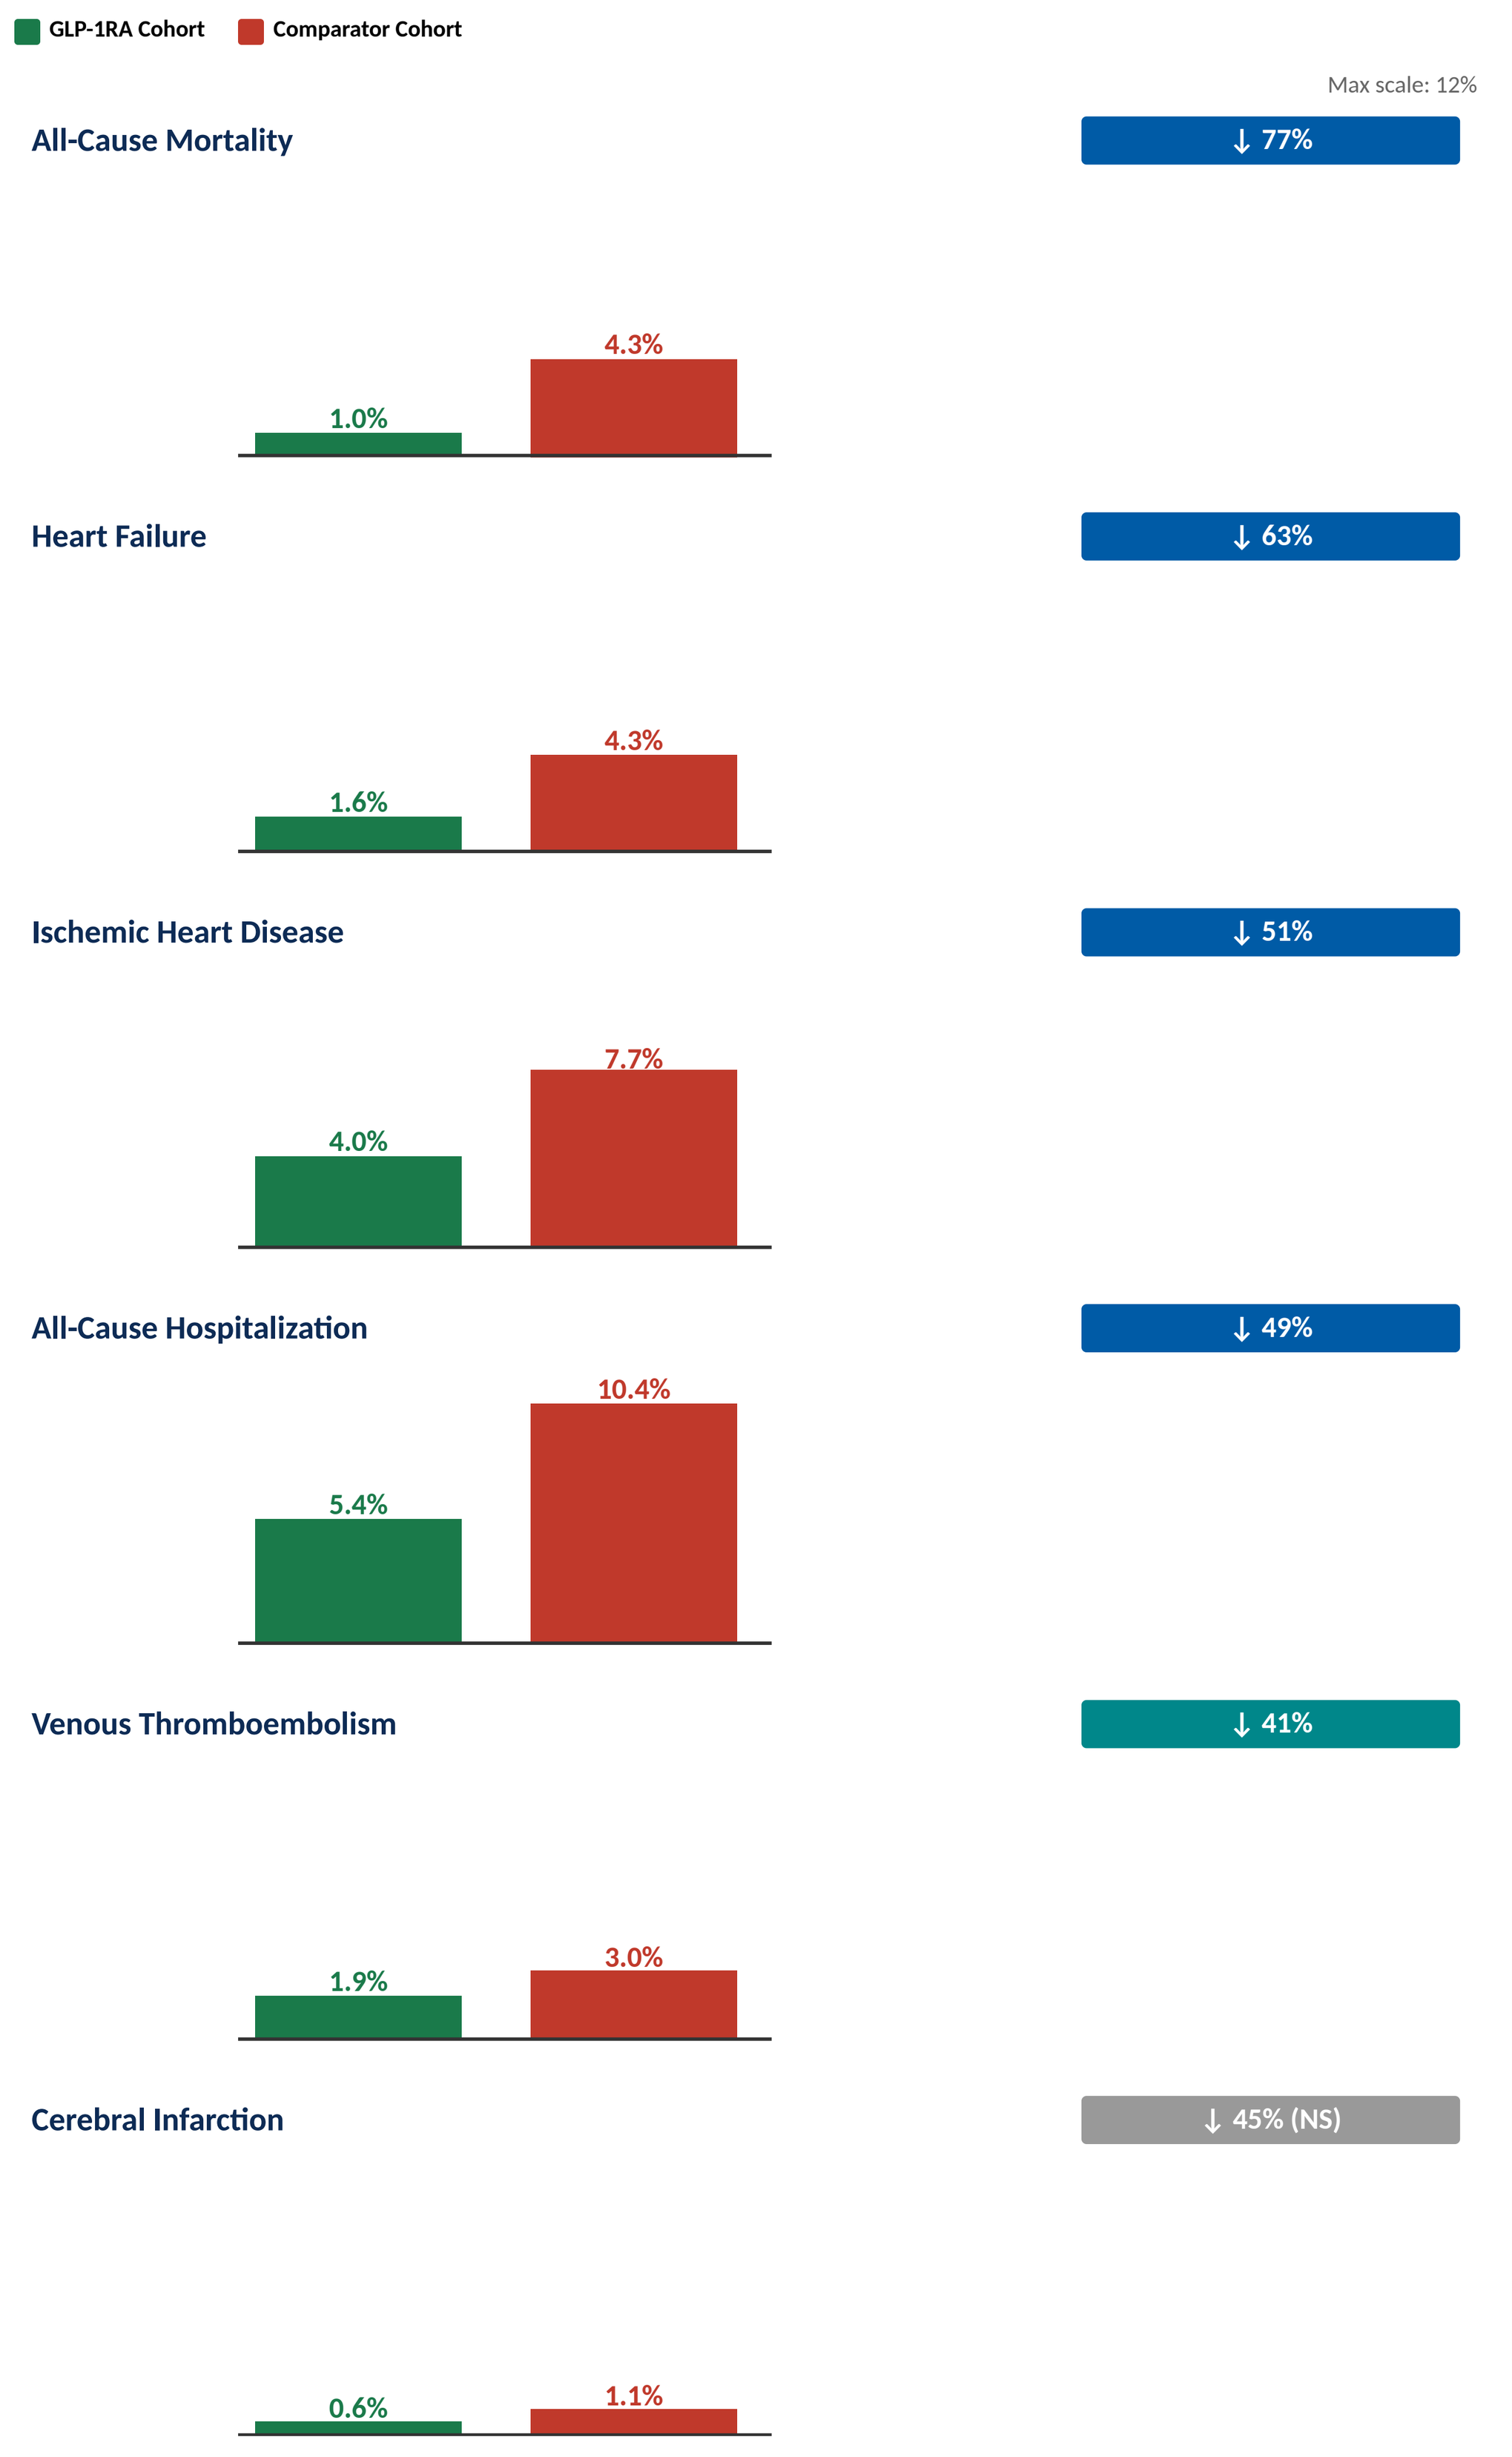
GLP-1RA Cohort
Comparator Cohort
Max scale: 12%
All-Cause Mortality
↓ 77%
1.0%
4.3%
Heart Failure
↓ 63%
1.6%
4.3%
Ischemic Heart Disease
↓ 51%
4.0%
7.7%
All-Cause Hospitalization
↓ 49%
5.4%
10.4%
Venous Thromboembolism
↓ 41%
1.9%
3.0%
Cerebral Infarction
↓ 45% (NS)
0.6%
1.1%

CONCLUSIONS

In this global PSM analysis of MPN patients with T2DM and/or obesity, GLP-1RA exposure was associated with significantly lower risks across multiple major outcomes:

- **↓ 77%** All-Cause Mortality — *HR 0.23 (p<0.001)*
- **↓ 63%** Heart Failure — *HR 0.37 (p<0.001)*
- **↓ 51%** Ischemic Heart Disease — *HR 0.49 (p<0.001)*
- **↓ 49%** All-Cause Hospitalization — *HR 0.51 (p<0.001)*
- **↓ 41%** Venous Thromboembolism — *HR 0.59 (p=0.020)*
- **↓ 45%** Cerebral Infarction — *HR 0.55 (p=0.108, NS)*

GLP-1RAs represent a promising repurposing candidate to reduce nonmalignant morbidity in classical MPNs. Prospective studies incorporating biomarker- and thrombosis-focused endpoints are warranted.

REFERENCES

1. Marchioli R, et al. *N Engl J Med.* 2013;368:22–33.
2. Marso SP, et al. (LEADER). *N Engl J Med.* 2016;375:311–322.
3. Marso SP, et al. (SUSTAIN-6). *N Engl J Med.* 2016;375:1834–1844.
4. Tefferi A, Barbui T. *Am J Hematol.* 2023;98:354–377.
5. TriNetX Global Collaborative Network. trinetx.com

DISCLOSURES

COI: No relevant conflicts of interest declared.

Funding: No external funding received.

Data: TriNetX Global Network (de-identified EHR; IRB exemption applies).

CORRESPONDING AUTHOR

Muluken Megiso, MD ✉
Department of Internal Medicine
Jefferson Einstein Philadelphia Hospital
Philadelphia, PA, USA
muluken.megiso@jefferson.edu

© 2026 Jefferson Health / Thomas Jefferson University | All data de-identified | TriNetX Global Collaborative Network | N = 1,837 per cohort after propensity score matching

GLP-1 = Glucagon-like Peptide-1 | MPN = Myeloproliferative Neoplasm | VTE = Venous Thromboembolism | PSM = Propensity Score Matching | PV = Polycythemia Vera | ET = Essential Thrombocythemia | PMF = Primary Myelofibrosis