Phase 1 Study to Evaluate Safety, Tolerability, Pharmacokinetics (PK), and Clinical Activity of the Antibody–Drug Conjugate (ADC) ARC-02 as Monotherapy and in Combination With Rituximab in Adults With B-Cell Non-Hodgkin Lymphoma (NHL)

Navid Hafez,[1] Eliza Hawkes,[2] Michael Gilbertson,[3] Tara Cochrane,[4] Katharine L. Lewis,[5,6] Sara Soukup,[7] Shinya Okamura,[7] Qing Xu,[7] Angela Hu,[7] Bernd Schlereth,[8] Vedran Radojcic,[7] Don Stevens,[9] Arvind Chaudry[10]

[1]START Los Angeles, Los Angeles, CA, USA; [2]Olivia Newton John Cancer Research Institute at Austin Health, Heidelberg, Australia; [3]Monash Health, Clayton, Australia; [4]Gold Coast University Hospital, Southport, Australia; [5]Department of Haematology, Sir Charles Gairdner Hospital, Nedlands, WA, Australia; [6]Linear Clinical Research, Nedlands, WA, Australia; [7]Taiho Oncology, Inc., Princeton, NJ, USA; [8]Araris Biotech AG, Au ZH, Switzerland; [9]Norton Cancer Institute, Louisville, KY, USA; [10]Summit Cancer Center, Spokane Valley, WA, USA

Introduction

- As many as 50% of individuals with B-cell non-Hodgkin lymphoma (NHL) may experience refractory or relapsed (R/R) disease, for which there are limited treatment options[1-3]
- Antibody–drug conjugates (ADC) are indicated for the treatment of aggressive high-grade lymphomas but are not approved for the treatment of other B-cell NHL subtypes. Toxicities induced by the unintended and premature release of the monomethyl auristatin E have limited the development of the anti-CD79b ADC polatuzumab-vedotin in other B-cell NHLs[4]
- ARC-02 is a novel anti-CD79b ADC developed using proprietary AraLinQ™ technology, which enhances ADC homogeneity and stability, offering improved on-target activity and enhanced safety
 - In preclinical studies, ARC-02 demonstrated a 16-fold higher therapeutic index over polatuzumab-vedotin (data on file). This was based on an up to 8-fold improvement in safety margin and potent cytotoxicity in NHL cell lines and xenograft models, with ARC-02 achieving comparable efficacy at 2-fold lower payload delivery.[5,6] Preclinical evidence also shows potential synergy between CD79b-targeted therapy and rituximab[7]

Study Design

- ARC-02-101 (NCT07691606) is a first-in-human open-label, dose-finding phase 1 study designed to evaluate the safety, tolerability, PK, pharmacodynamics, and preliminary clinical activity of ARC-02 alone and in combination with rituximab in adults with R/R B-cell NHL
- The study consists of Part A (ARC-02 monotherapy) and Part B (ARC-02 in combination with rituximab) (**Figure 1**). Both Part A and Part B include a dose escalation phase followed by a dose expansion phase. Commencement of Part B will be informed by findings from Part A dose escalation
- Approximately 140 participants are planned to be enrolled in both monotherapy and combination therapy cohorts in Part A and Part B
- In Part A (monotherapy), dose escalation follows accelerated titration design with single-participant cohorts used for the first 2 dose levels, before transitioning to a Bayesian Optimal Interval (BOIN) design. BOIN methodology will be used to estimate the maximum tolerated dose and is defined as the highest dose level with a dose-limiting toxicity (DLT) rate lower than the prespecified target toxicity rate of 25%. Approximately 100 participants will be enrolled in Part A (dose escalation will enroll up to 12 participants per dose level [approx. n=40] and dose expansion will enroll up to 20 participants per disease cohort (indolent NHL [iNHL], diffuse large B-cell NHL, mantle cell lymphoma; [approx. n=60])
- Part B (ARC-02 combination with rituximab) will initiate after the ARC-02 monotherapy maximum safely administered dose (MSAD) has been determined in Part A and will enroll approximately 40 participants
 - In the dose escalation portion, the starting dose (one dose level below the ARC-02 monotherapy MSAD) and the ARC-02 monotherapy MSAD dose will be evaluated
 - The dose expansion portion will enroll up to 20 participants with iNHL (follicular lymphoma and marginal zone lymphoma). Data from this cohort will inform the recommended phase 2 dose (RP2D) for ARC-02 in combination with rituximab. The Lugano criteria[8] will be used to assess response in participants with radiographically measurable disease

Study Status

- **Study start date:** June 22, 2026
- **Estimated study completion date:** December 2030
- As of August 26, 2026, two participants have been enrolled

Figure 1: Study design

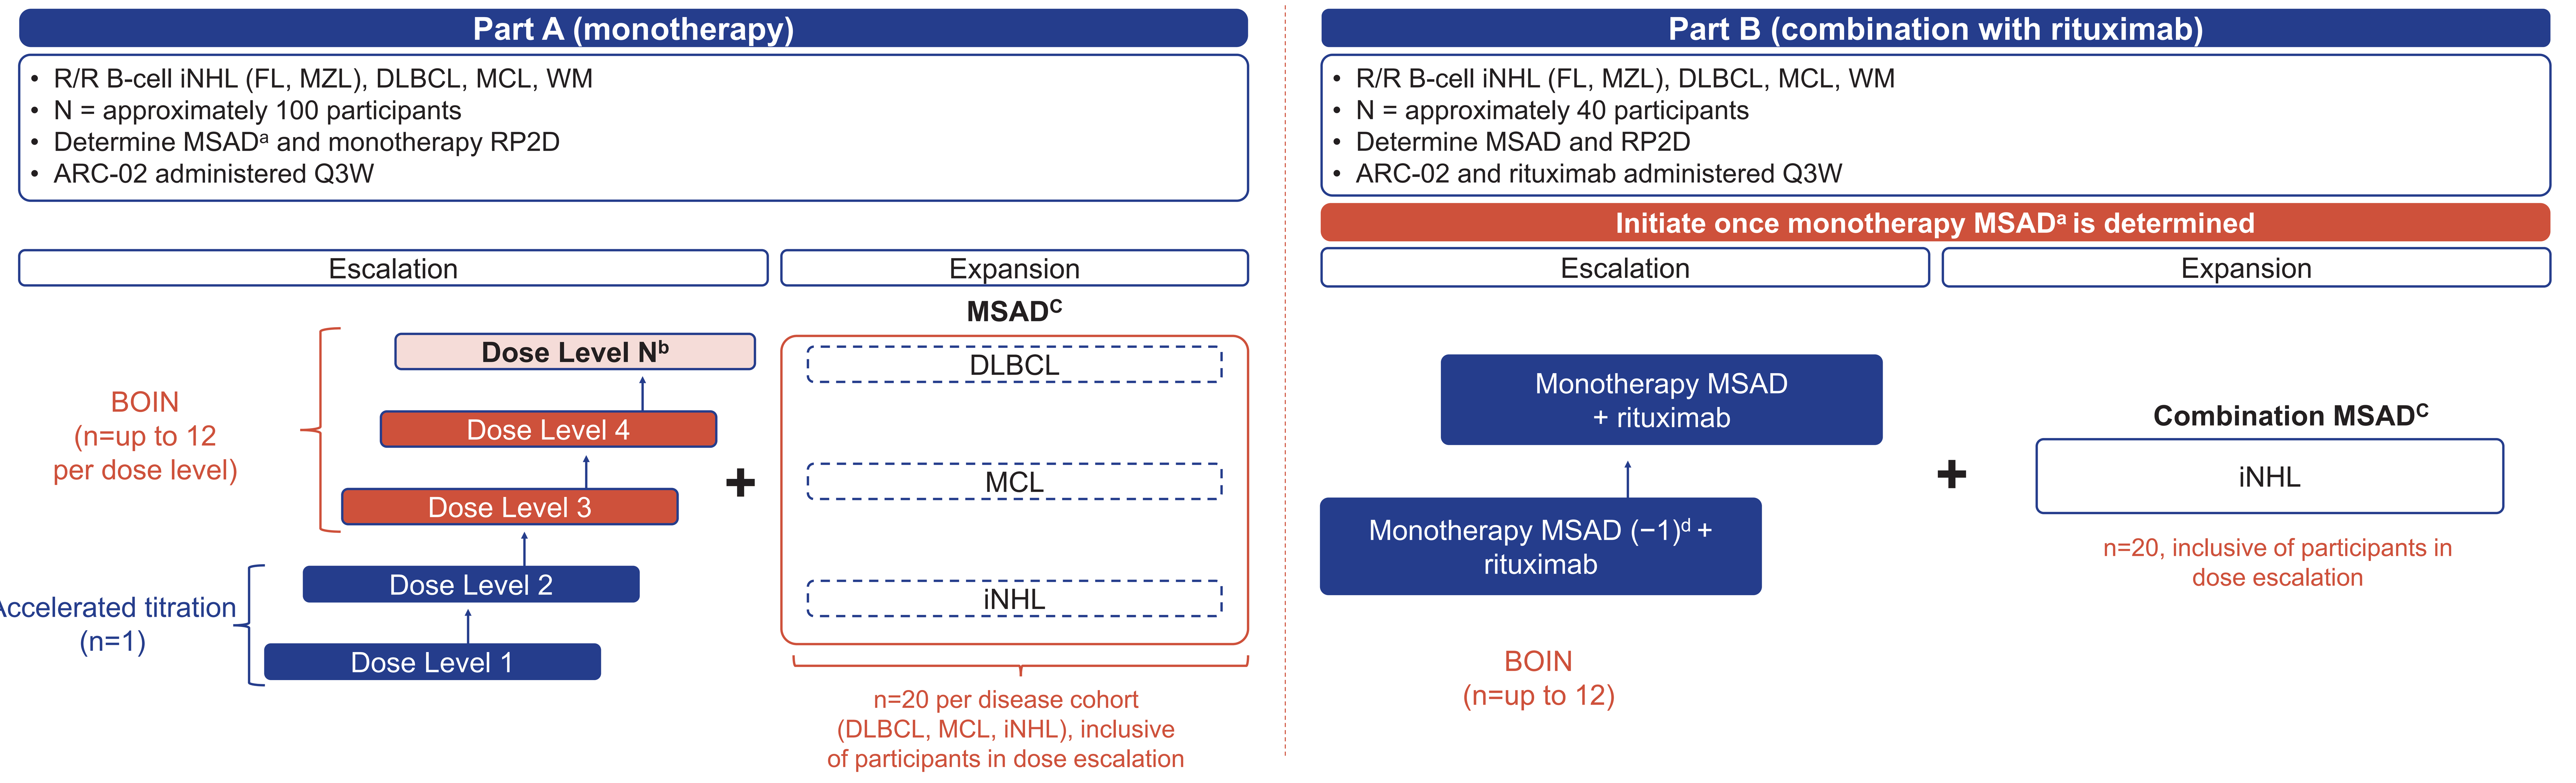
Part A (monotherapy)
R/R B-cell iNHL (FL, MZL), DLBCL, MCL, WM
N = approximately 100 participants
Determine MSADa and monotherapy RP2D
ARC-02 administered Q3W
Escalation
Expansion
MSADc
Dose Level Nb
Dose Level 4
Dose Level 3
Dose Level 2
Dose Level 1
BOIN (n=up to 12 per dose level)
Accelerated titration (n=1)
DLBCL
MCL
iNHL
n=20 per disease cohort (DLBCL, MCL, iNHL), inclusive of participants in dose escalation
Part B (combination with rituximab)
R/R B-cell iNHL (FL, MZL), DLBCL, MCL, WM
N = approximately 40 participants
Determine MSAD and RP2D
ARC-02 and rituximab administered Q3W
Initiate once monotherapy MSADa is determined
Escalation
Expansion
Monotherapy MSAD + rituximab
Monotherapy MSAD (−1)d + rituximab
BOIN (n=up to 12)
Combination MSADc
iNHL
n=20, inclusive of participants in dose escalation

[a]MSAD represents the maximum safely administered dose.
[b]Dose levels above Dose Level 4 may be evaluated if supported when sufficient clinical data are available at the lower dose levels and after protocol amendment.
[c]If results of expansion cohort justify further ARC-02 development, to support dose optimization, an additional lower dose level (per B-cell NHL type) may be expanded after protocol amendment.
[d](−1) represents a dose level lower than the monotherapy MSAD.
BOIN, Bayesian Optimal Interval; DLBCL, diffuse large B-cell lymphoma; FL, follicular lymphoma; iNHL, indolent non-Hodgkin lymphoma; MCL, mantle cell lymphoma; MSAD, maximum safely administered dose; MZL, marginal zone lymphoma; NHL, non-Hodgkin lymphoma; Q3W, every 3 weeks; RP2D, recommended phase 2 dose; R/R, relapsed/refractory; WM, Waldenstrom macroglobulinemia.

Participant Eligibility Criteria

Key inclusion criteria	Key exclusion criteria
• Aged ≥18 years	• Grade >1 neuropathy
• B-cell NHL with histology based on WHO criteria[9] and disease requiring treatment: - FL grades 1–3B - MZL - MCL - DLBCL - Other B-cell NHL	• Has any of the following medical conditions: - Active infection - Active ILD/pneumonitis or history of ILD/pneumonitis requiring treatment - Concurrent malignancy (progressing or requires treatment) - Active CNS lymphoma involvement requiring treatment - Cardiac event within 6 months prior to study entry - QTcF >470 ms
• Measurable disease, defined by Lugano classification	• Has received chemotherapy, radiotherapy, small molecule, investigational, and biologic agents (including CD79b-directed agents) within 28 days (or at least 5 half-lives, whichever is shorter), prior to the first dose of the study intervention
• Has received at least 2 prior lines of systemic therapies for B-cell NHL and is not eligible to receive additional standard of care therapies	• Use of any live or live-attenuated vaccine within 28 days before the first dose of the study intervention
• ECOG performance status of ≤2	• Use of immunosuppression for management of allogeneic HCT within 3 months prior to the first dose of the study intervention
• Adequate organ function	• CAR-T or autologous HCT within 100 days prior to the first dose of the study intervention
	• Ongoing toxicity from prior anticancer therapy that has not resolved[a]

[a]Resolved to grade ≤2 (neutropenia) or grade ≤1 (thrombocytopenia or nonhematologic toxicities, with the exception of alopecia).
CAR-T, chimeric antigen receptor T-cell; CNS, central nervous system; DLBCL, diffuse large B-cell lymphoma; ECOG, Eastern Cooperative Oncology Group; FL, follicular lymphoma; HCT, hematopoietic cell transplantation; ILD, interstitial lung disease; MCL, mantle cell lymphoma; MZL, marginal zone lymphoma; NHL, non-Hodgkin lymphoma; WHO, World Health Organization.

Endpoints

Endpoint	Dose escalation	Dose expansion
Primary	• Incidence of DLTs and TEAEs (including SAEs)	• ORR (investigator-assessed)
Secondary	• ORR (investigator-assessed)	• Number of participants with TEAEs and SAEs
	• TTR • DoR • PFS • PK parameters for acMMAE, total anti-CD79b antibody and free MMAE (C_{max}, $T_{½}$, $AUC_{0\text{-}T}$, and $AUC_{0\text{-}\infty}$) • Presence of ADA and nAb	

acMMAE, antibody-conjugated monomethyl auristatin E; ADA, antidrug antibody; $AUC_{0\text{-}T}$, area under the curve from time to last measurable concentration; $AUC_{0\text{-}\infty}$, area under the curve from time 0 to infinity; C_{max}, maximum serum concentration; DLT, dose-limiting toxicity; DoR, duration of response; MMAE, monomethyl auristatin E; nAb, neutralizing antibody; ORR, objective response rate; PFS, progression-free survival; PK, pharmacokinetics; SAE, serious adverse event; $T_{½}$, half-life; TEAE, treatment-emergent adverse event; TTR, time to response.

References

1. Crump M, et al. *Blood*. 2017;130:1800–1808.
2. Sehn LH, et al. *Blood*. 2007;109:1857–1861.
3. Sehn LH, Salles G. *N Engl J Med*. 2021;384:842–858.
4. Polivy US Prescribing Information. March 2026
5. Schlereth B, et al. *Blood*. 2022;140(Supplement 1):11568–11569.2022
6. Attinger-Toller I, et al. *Cancer Res*. 2022;82(Supplement 12):2910.
7. Kawasaki N, et al. *Br J Haematol*.2022;199:245–255.
8. Cheson BD, et al. *J Clin Oncol*. 2014;32:3059–3067.
9. Swerdlow S, et al. *Blood*. 2016;127:2375–2387.

Acknowledgements

This study is sponsored by Taiho Oncology, Inc. Medical writing assistance was provided by Kerry K. Brinkman, PhD, from Envision Pharma Group, funded by Taiho Oncology, Inc.

Presented at the SOHO Annual Meeting 2026; September 9–12, 2026; Houston, TX (ABCL - 1058)