IBCL - 334

Fixed Duration Subcutaneous Mosunetuzumab had Better Practice Efficiency versus Treat-to-Progression Subcutaneous Epcoritamab in Third-Line or Later Follicular Lymphoma: An Economic Analysis

Zahra Mahmoudjafari,[1] Katherine L. Rosettie,[2] Mei Wu,[2]
Shih-Wen Lin,[2]* Carolina Reyes,[2] Esprit Ma[2]
**e-mail: lins31@gene.com*

Summary

Data on the healthcare professionals (HCPs) involved in administering subcutaneous (SC) bispecific antibodies and the associated time commitments for HCPs and patients are limited

The practice efficiency of fixed duration mosunetuzumab SC and treat-to-progression epcoritamab SC in third-line or later (3L+) follicular lymphoma (FL) is presented

In patients with 3L+ FL, fixed duration mosunetuzumab SC, with fewer doses of corticosteroid premedications and treatment doses versus treat-to-progression epcoritamab SC, demonstrated better practice efficiency, saving 42 hours of HCP time and 31 hours of patient time compared with epcoritamab SC

[1]The University of Kansas Health System, Kansas City, KS, USA;
[2]Genentech, Inc., South San Francisco, CA, USA.

Background

- The SC formulation of mosunetuzumab was approved by the United States (US) Food and Drug Administration in December 2025 for the treatment of patients with relapsed or refractory FL after two or more lines of therapy, based on the Phase II study of mosunetuzumab SC demonstrating non-inferior pharmacokinetics compared with intravenous mosunetuzumab.[1,2]
- However, data on the HCPs involved in administering SC bispecific antibodies and the associated time commitments for HCPs and patients are limited.
- The following economic analysis was conducted to estimate the practice efficiency of fixed duration mosunetuzumab SC and treat-to-progression epcoritamab SC in 3L+ FL over 1 year, from a US health systems perspective.

Methods

- Differences in practice efficiency between mosunetuzumab SC and epcoritamab SC were evaluated from before to during bispecific antibody administration; routine care for post-bispecific antibody administration was excluded as it was assumed to be the same regardless of the bispecific antibody option (**Figure 1**).
- Data on corticosteroid premedication requirements, bispecific antibody dosing schedules and mean duration of treatment (DOT) were based on respective pivotal Phase I/II trials (mosunetuzumab SC: NCT02500407; epcoritamab SC: NCT03625037) and US prescribing labels.[2–5]
- Corticosteroid premedications before bispecific antibody administration were assumed to be administered by HCPs only.
- Corticosteroids on subsequent days for epcoritamab were assumed to be dispensed through a one-time retail pharmacy visit by patients
 - Clinical experts validated the time estimate (0.4 hours) for patients to do a round-trip drive to a retail pharmacy, based on the 5-mile distance from residence to retail pharmacy in the US reported by Berenbrok, et al. 2022,[6] at a speed limit of 25 miles per hour in urban areas.
- Information on HCPs involved and mean time spent by HCPs and patients was based on input by US-based clinicians
 - Average HCP time before bispecific antibody administration was calculated from the time spent on laboratory tests, premedications, and drug preparation and verification
 - Average HCP time during and average patient time before bispecific antibody administration were both calculated from the time spent on laboratory tests and corticosteroid premedications
 - Based on clinical input, the average patient time during bispecific antibody administration was assumed to be the same in Cycle 1 and Cycle 2+.
- Costs were estimated using mean hourly wages from the US Bureau of Labor Statistics, adjusted to 2026 US dollars.[7]

Figure 1. Model framework.

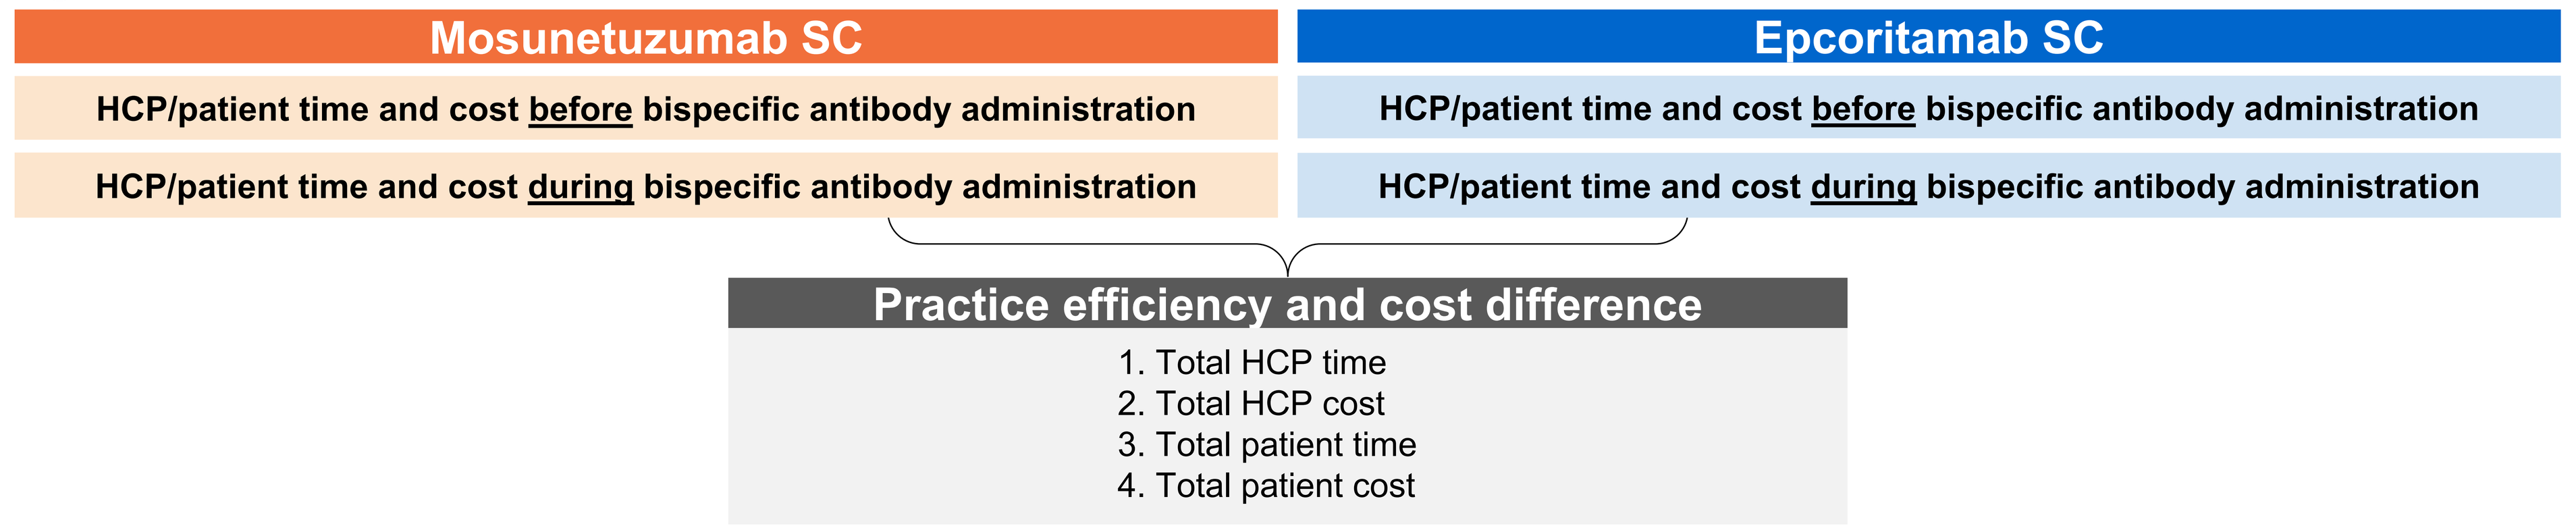

Mosunetuzumab SC
HCP/patient time and cost before bispecific antibody administration
HCP/patient time and cost during bispecific antibody administration
Epcoritamab SC
HCP/patient time and cost before bispecific antibody administration
HCP/patient time and cost during bispecific antibody administration
Practice efficiency and cost difference
1. Total HCP time
2. Total HCP cost
3. Total patient time
4. Total patient cost

Results

- HCPs involved in drug preparation and verification comprised pharmacy technicians, pharmacists, and registered nurses.
- HCPs involved in patient visits comprised front desk staff, laboratory technicians, pharmacy technicians, pharmacists, registered nurses, medical assistants, physician assistants, and nurse practitioners.
- Over 1 year, patients receiving mosunetuzumab SC required fewer doses of corticosteroid premedications (three times before weekly mosunetuzumab in Cycle 1) versus epcoritamab SC (16 times, before and after weekly epcoritamab in Cycle 1) and fewer doses (10 doses for patients achieving complete response by Cycle 8 or at most 19 doses) versus epcoritamab SC (28 doses) (**Figure 2**).
- At the respective mean DOTs, fewer bispecific antibody administrations were required with mosunetuzumab SC versus epcoritamab SC (**Figure 2**).

Figure 2. Corticosteroid premedications and dosing schedules for mosunetuzumab SC and epcoritamab SC.

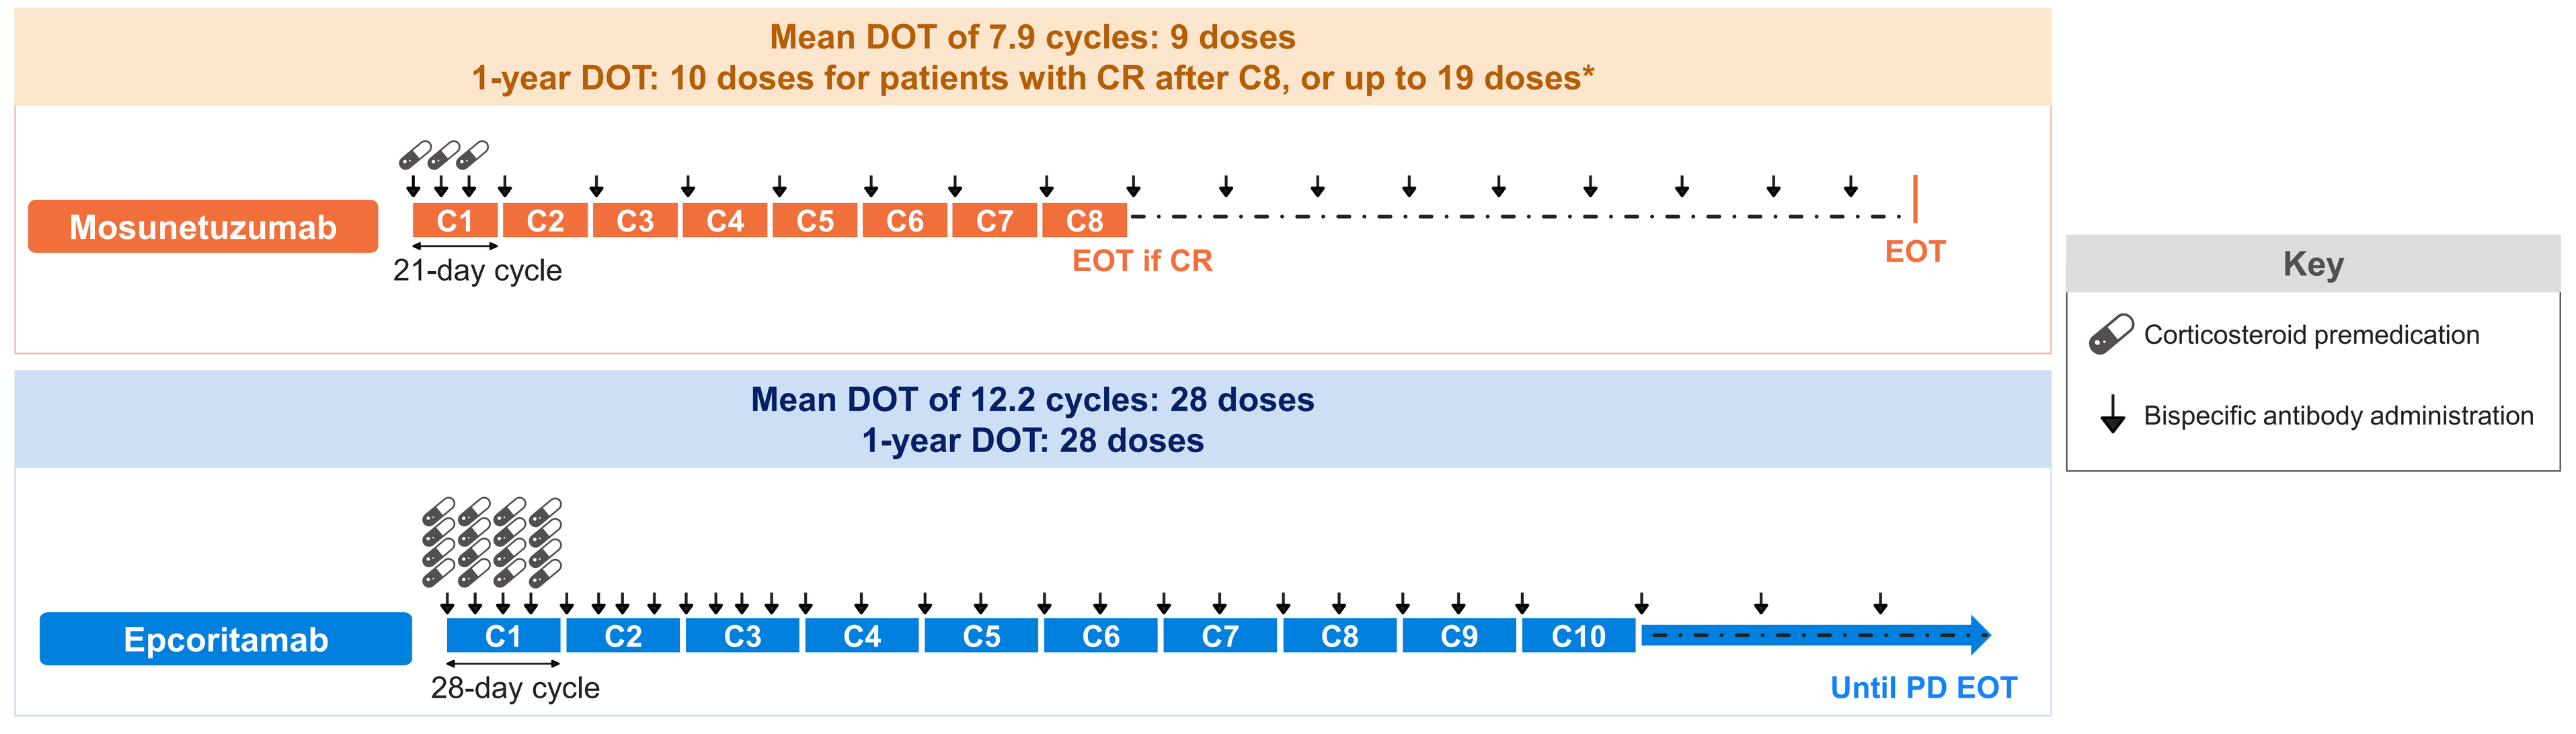

Mean DOT of 7.9 cycles: 9 doses
1-year DOT: 10 doses for patients with CR after C8, or up to 19 doses*
Mosunetuzumab
C1 C2 C3 C4 C5 C6 C7 C8
21-day cycle
EOT if CR
EOT
Mean DOT of 12.2 cycles: 28 doses
1-year DOT: 28 doses
Epcoritamab
C1 C2 C3 C4 C5 C6 C7 C8 C9 C10
28-day cycle
Until PD EOT
Key
Corticosteroid premedication
Bispecific antibody administration

*Up to 19 doses for patients with a partial response or stable disease after C8 (not in patients with PD).
C, Cycle; CR, complete response; EOT, end of treatment; PD, progressive disease.

Results (continued)

- Fixed duration mosunetuzumab SC had numerically better practice efficiency, with a total of 42 HCP hours (**Figure 3A**) and 31 patient hours (**Figure 3B**) saved compared with treat-to-progression epcoritamab SC in patients with 3L+ FL.

Figure 3. Total HCP (A) and patient (B) time at mean DOT for mosunetuzumab SC and epcoritamab SC.

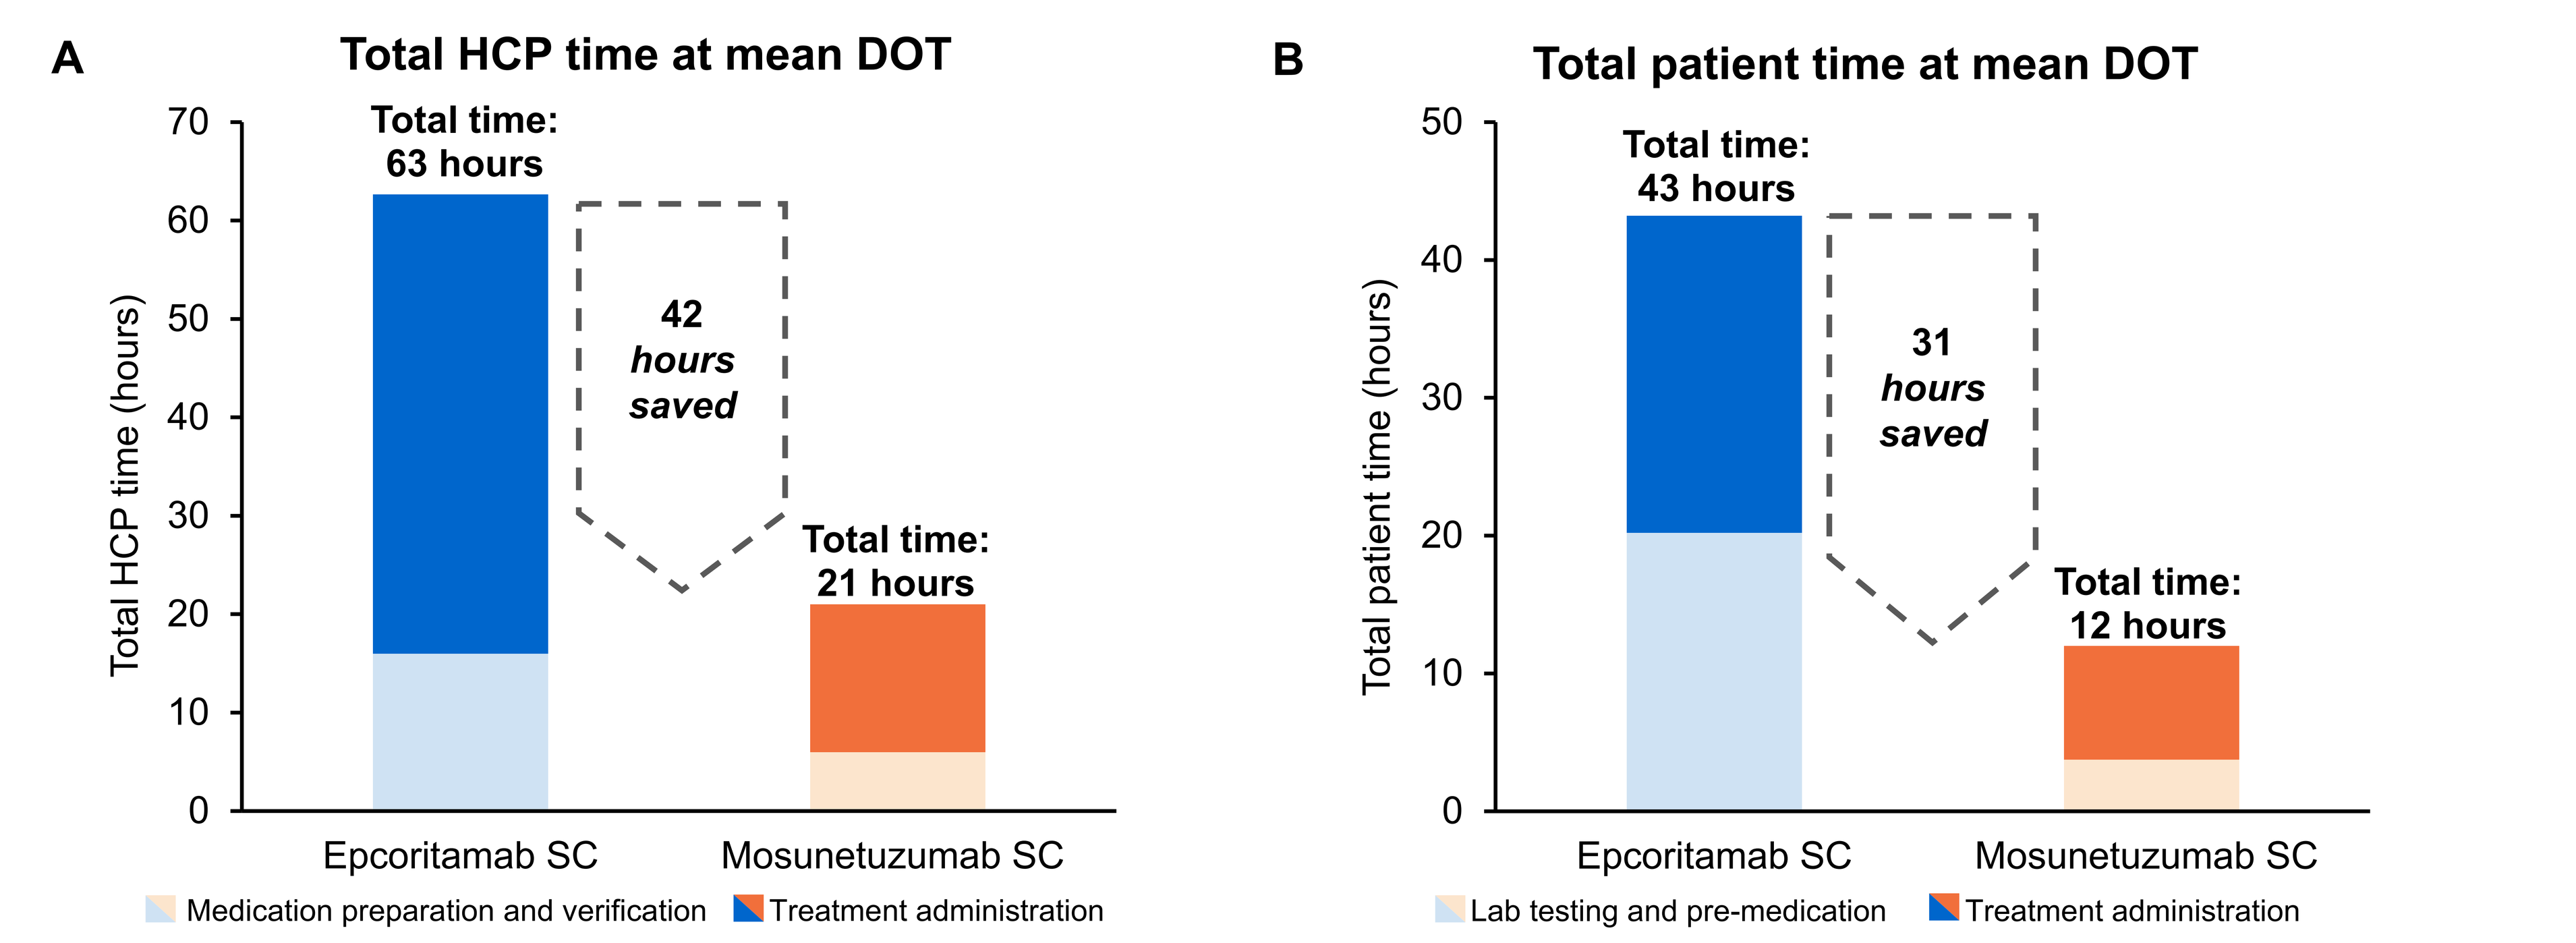

A
Total HCP time at mean DOT
Total HCP time (hours)
0 10 20 30 40 50 60 70
Total time: 63 hours
42 hours saved
Total time: 21 hours
Epcoritamab SC
Mosunetuzumab SC
Medication preparation and verification
Treatment administration
B
Total patient time at mean DOT
Total patient time (hours)
0 10 20 30 40 50
Total time: 43 hours
31 hours saved
Total time: 12 hours
Epcoritamab SC
Mosunetuzumab SC
Lab testing and pre-medication
Treatment administration

- The HCP and patient hours saved with mosunetuzumab SC versus epcoritamab SC resulted in cost savings of $1,972 (**Figure 4A**) and $1,160 (**Figure 4B**), respectively.

Figure 4. Total HCP (A) and patient (B) costs at mean DOT for mosunetuzumab SC and epcoritamab SC.

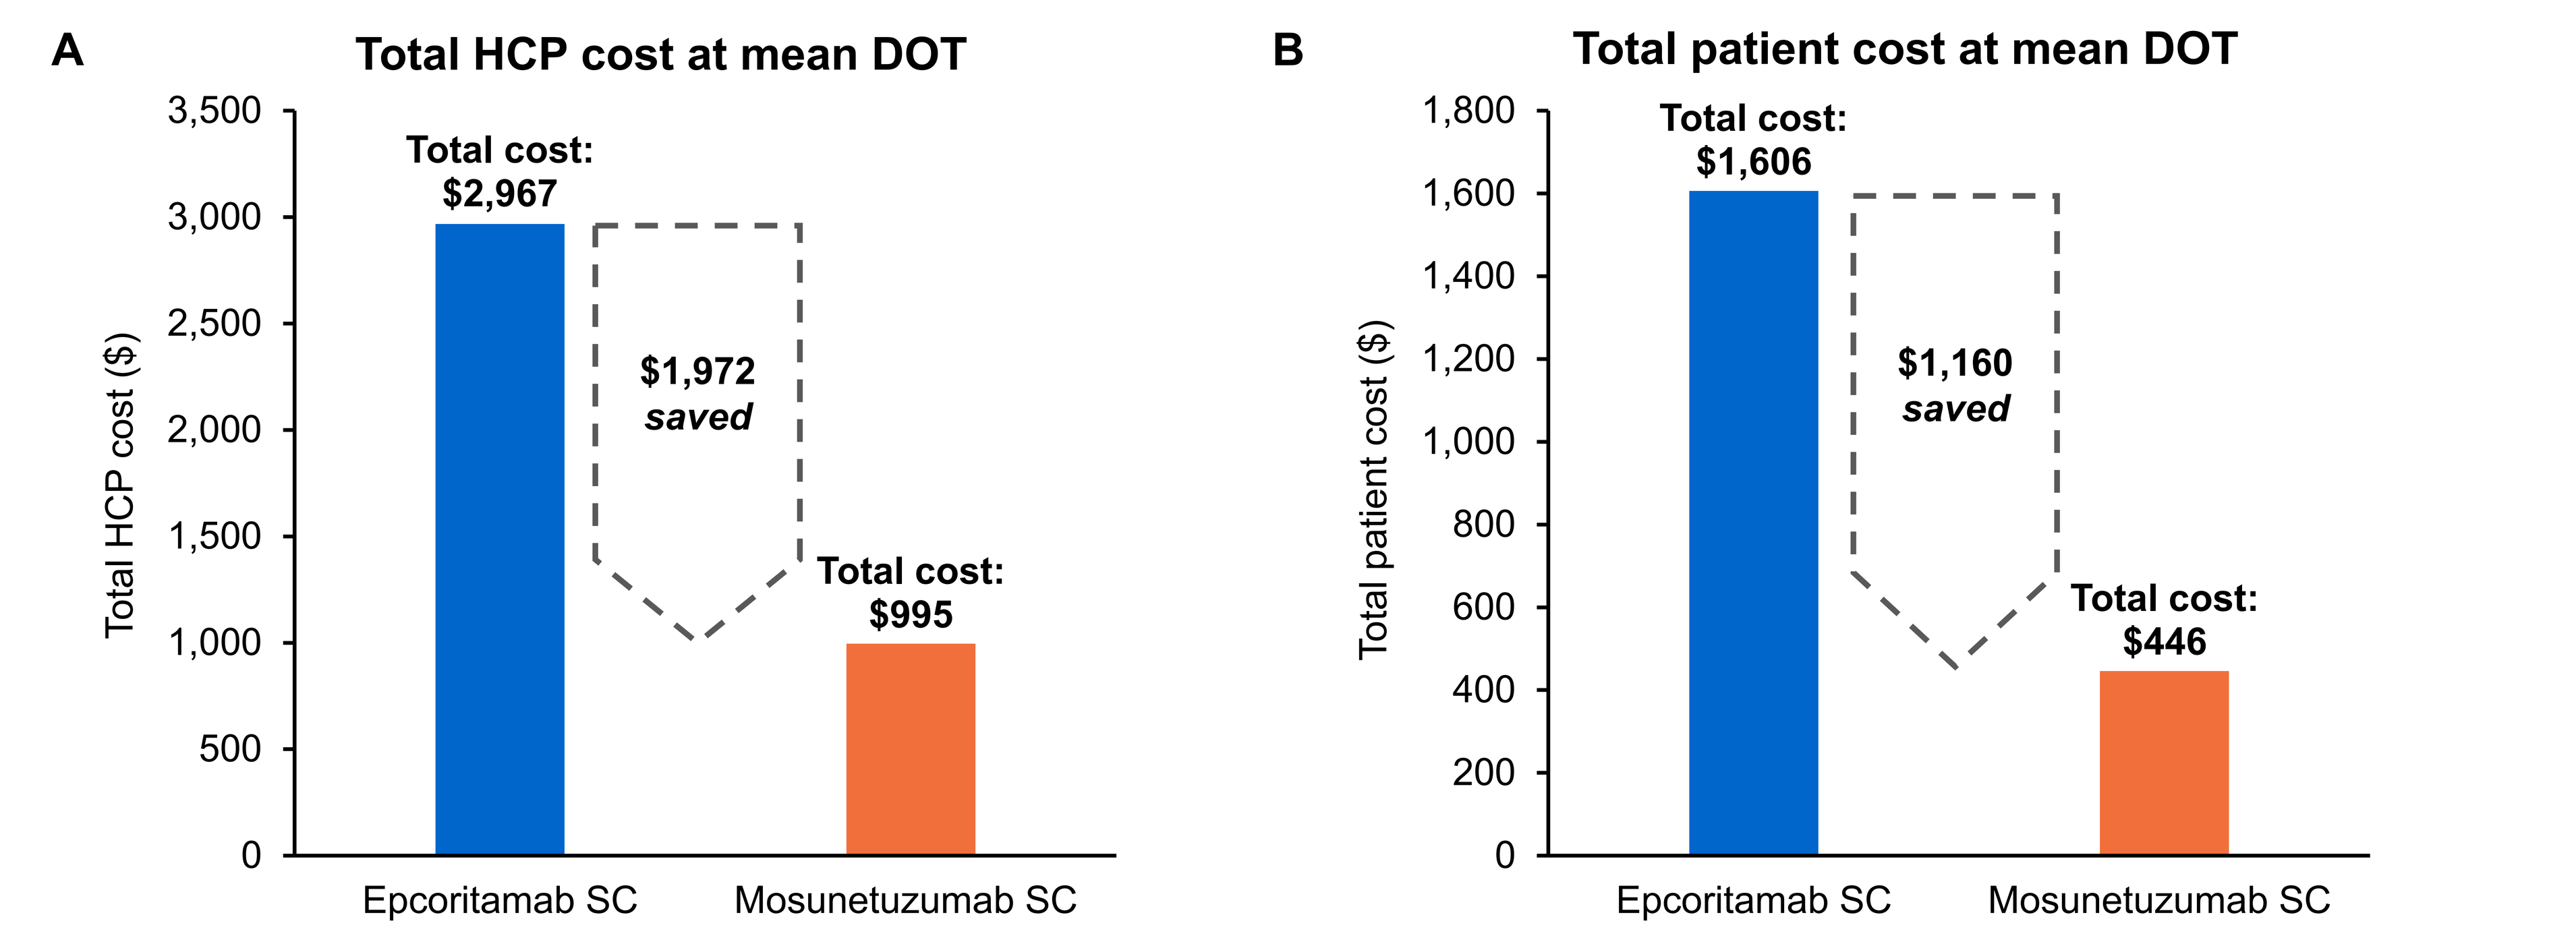

A
Total HCP cost at mean DOT
Total HCP cost ($)
0 500 1,000 1,500 2,000 2,500 3,000 3,500
Total cost: $2,967
$1,972 saved
Total cost: $995
Epcoritamab SC
Mosunetuzumab SC
B
Total patient cost at mean DOT
Total patient cost ($)
0 200 400 600 800 1,000 1,200 1,400 1,600 1,800
Total cost: $1,606
$1,160 saved
Total cost: $446
Epcoritamab SC
Mosunetuzumab SC

Limitations

- This analysis focused on the practice efficiency of SC bispecific antibody administration; neither safety nor efficacy data were included in the analysis.
- For mosunetuzumab SC, the total number of doses needed was calculated using the mean DOT reported in its pivotal trial; however, the DOT may vary in routine clinical settings.
- For epcoritamab SC, the mean DOT was extrapolated from the median DOT reported in the 3-year follow-up of the pivotal trial,[8] using the method described by Hozo, et al. 2025.[9] However, the DOT may differ in routine clinical settings.
- The average time estimates for each unit were based on the clinical input from a single US institution and therefore, may not be representative of all practices (e.g., academic versus community) or workflows across the US.
- The assumption that patients would travel to a retail pharmacy for subsequent epcoritamab corticosteroid premedication was based on clinical input from one institution, and the average unit time estimate for this travel was derived from literature and validated by clinicians; therefore, these assumptions may not be generalizable to all patients in the US.

Conclusions

- In patients with 3L+ FL, fixed duration mosunetuzumab SC, with fewer doses of corticosteroid premedications and treatment doses versus treat-to-progression epcoritamab SC, demonstrated better practice efficiency, saving 42 hours of HCP time and 31 hours of patient time compared with epcoritamab SC.
- Reducing the time associated with receiving treatment may lessen the burden of treatment, giving patients and their caregivers more time for daily activities and potentially improving overall quality of life. It may also improve healthcare resource utilization by freeing up clinical capacity, enabling healthcare professionals to focus on other critical aspects of patient care.
- This analysis offers valuable insights for optimizing the use of SC bispecific antibodies in patients with 3L+ FL and for informing shared decision-making between HCPs and patients regarding bispecific antibody treatment selection.

Presented at the 14th Annual Meeting of the Society of Hematologic Oncology (SOHO 2026) | September 9–12, 2026

References

1. Bartlett NL, et al. Blood. 2024;144:1645–7.
2. LUNSUMIO VELO USPI. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2025/761263s006lbl.pdf. Accessed July 2026.
3. EPKINLY USPI. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/761324s000lbl.pdf. Accessed July 2026.
4. Bartlett NL, et al. Am J Hematol. 2026;101:986–97.
5. Linton KM, et al. Lancet Haematol. 2024;11:e593–605.
6. Berenbrok LA, et al. JAPhA. 2022;62:1816–22.
7. US Bureau of Labor Statistics. Available at: https://www.bls.gov/home.htm. Accessed July 2026.
8. Vitolo U, et al. EHA. 2025. PF881.
9. Hozo SP, et al. BMC Med Res Methodol. 2005;5:13.

Acknowledgments

This study was sponsored by Genentech, Inc. Third-party editorial assistance, under the direction of authors, was provided by Berta Fatás, PhD, and Anna Ditchfield, MChem, of Ashfield MedComms, an Inizio company, and was funded by Genentech, Inc.

Disclosures

ZM: Honoraria, consulting or advisory role (paid to author): Pfizer, Sanofi, Genmab, Genentech, Inc., BMS. **KLR**: Current employment: Genentech, Inc.; Stock or other ownership (paid to author): F. Hoffmann-La Roche Ltd. **MW**: Current employment: Genentech, Inc.; Stock or other ownership (paid to author): F. Hoffmann-La Roche Ltd. **SL**: Current employment: Genentech, Inc.; Stock or other ownership (paid to author): F. Hoffmann-La Roche Ltd. **CR**: Current employment: Genentech, Inc.; Stock or other ownership (paid to author): F. Hoffmann-La Roche Ltd; Research funding (paid to author): Genentech, Inc.; Travel, accommodations, expenses (paid to author): Genentech, Inc. **EM**: Current employment: Genentech, Inc.; Stock or other ownership (paid to author): F. Hoffmann-La Roche Ltd.