MM-134

Durable clinical benefit with BCMA-directed therapy, belantamab mafodotin plus pomalidomide and dexamethasone in relapsed/refractory multiple myeloma: DREAMM-8 long-term responder analysis

Meletios A. Dimopoulos,[1,2] Meral Beksaç,[3] Sosana Delimpasi,[4] Vladimir Vorobyev,[5] Hang Quach,[6] Ivan Špička,[7] Jakub Radocha,[8] Paweł Robak,[9] Kihyun Kim,[10] Syed Zafar,[11] Ainslee Moore,[12] Kristin Morris,[13] Margaret Polinkovsky,[14] Kelly Brock,[14] Joanna Grams,[15] Elisabet E. Manasanch,[14,a] María-Victoria Mateos,[16] Suzanne Trudel[17]

[a] Institution where work was conducted.
[1]Department of Clinical Therapeutics, School of Medicine, National and Kapodistrian University of Athens, Athens, Greece; [2]Department of Medicine, Korea University, Seoul, Republic of Korea; [3]Department of Hematology, Ankara Liv Hospital, Istinye University, Ankara, Turkey; [4]General Hospital Evangelismos, Athens, Greece; [5]Leningrad Regional Clinical Hospital, Saint Petersburg, Russian Federation; [6]University of Melbourne, St Vincent's Hospital Melbourne, Melbourne, VIC, Australia; [7]First Faculty of Medicine, Charles University and General Hospital, Prague, Czech Republic; [8]4th Department of Internal Medicine – Hematology, University Hospital Hradec Králové, Charles University, Faculty of Medicine in Hradec Králové, Hradec Králové, Czech Republic; [9]Medical University of Łódź, Łódź, Poland; [10]Sungkyunkwan University, Samsung Medical Center, Seoul, Republic of Korea; [11]Florida Cancer Specialists and Research Institute, Fort Myers, FL, USA; [12]GSK, Stevenage, UK; [13]GSK, Durham, NC, USA; [14]GSK, Collegeville, PA, USA; [15]GSK, Warsaw, Poland; [16]Hematology Department, University Hospital of Salamanca/IBSAL/Cancer Research Center-IBMCC (USAL-CSIC), Salamanca, Spain; [17]Department of Medical Oncology and Hematology, Princess Margaret Cancer Centre, Toronto, ON, Canada

Almost 40% of patients treated with BPd had prolonged remission as long-term responders (PFS ≥30 months), and >80% of these patients maintained PFS at 4 years

Digital poster

SCAN ME

Background

- Targeting BCMA has transformed the RRMM treatment landscape, delivering unprecedented survival benefits over standard-of-care triplets[1,2]
- Belamaf is an antibody-drug conjugate representing a unique BCMA-targeting modality that is accessible across all sites of care
- Belamaf, in combination with Pd, significantly improved PFS vs PVd in the DREAMM-8 phase 3 trial (HR, 0.52; 95% CI, 0.37-0.73; *P*<0.001)[3]
- This post hoc analysis was conducted to investigate the characteristics of and outcomes in patients who achieved sustained clinical benefit with BPd and are considered LTRs

Conclusions

Almost 40% of patients treated with BPd achieved LTR status, representing a 2.5-fold higher rate than with PVd

Higher LTR rates were also seen with BPd in patients with high-risk cytogenetics

BPd-treated LTRs had deeper responses compared with PVd-treated LTRs

BPd-treated LTRs demonstrated a trend toward prolonged PFS and PFS2

The safety profile in LTRs (PFS ≥30 months) was generally consistent with previous reports in the overall safety population[3]

Methods

- DREAMM-8 is a phase 3 trial of BPd vs PVd in patients with RRMM after ≥1 prior LOT including lenalidomide[3]

DREAMM-8 study design[3]

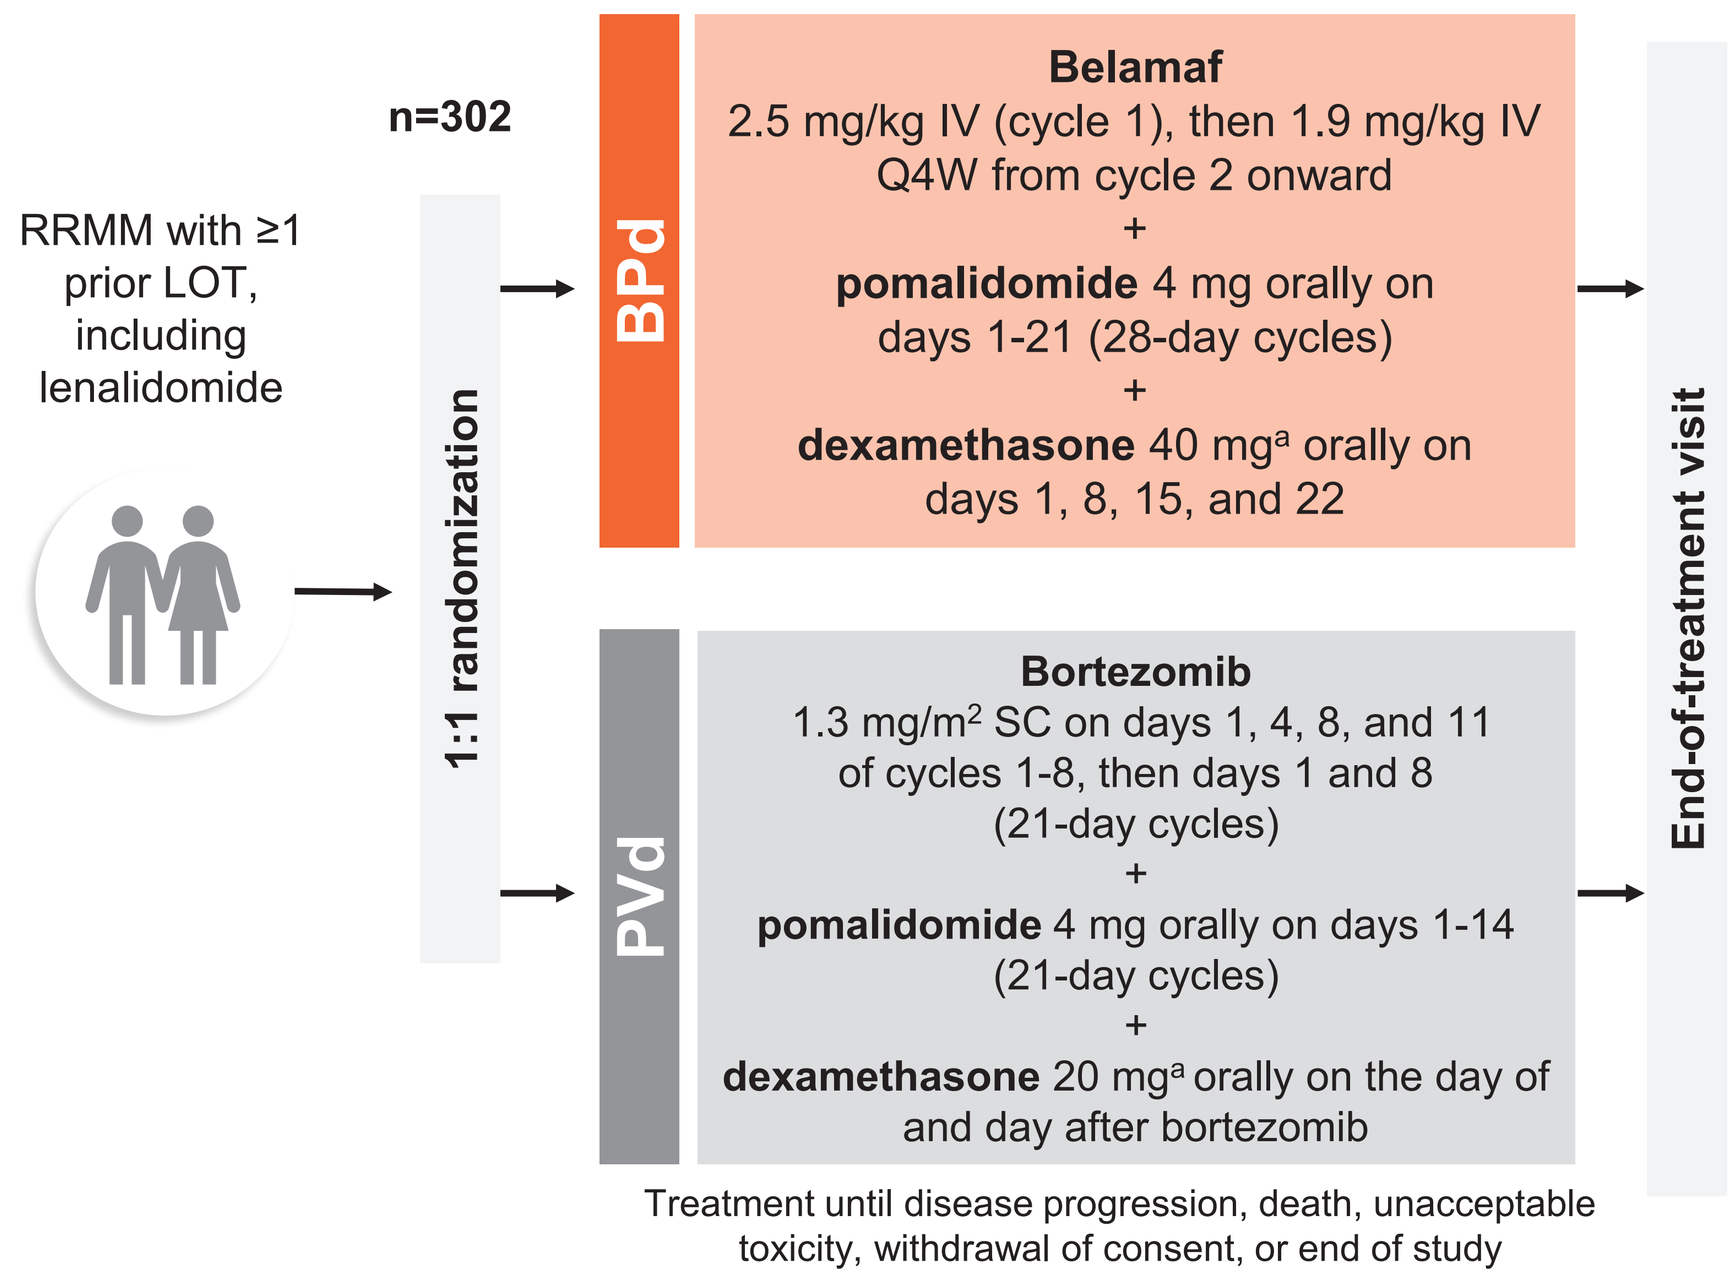

n=302
RRMM with ≥1 prior LOT, including lenalidomide
1:1 randomization
BPd
Belamaf
2.5 mg/kg IV (cycle 1), then 1.9 mg/kg IV Q4W from cycle 2 onward
+
pomalidomide 4 mg orally on days 1-21 (28-day cycles)
+
dexamethasone 40 mg[a] orally on days 1, 8, 15, and 22
PVd
Bortezomib
1.3 mg/m² SC on days 1, 4, 8, and 11 of cycles 1-8, then days 1 and 8 (21-day cycles)
+
pomalidomide 4 mg orally on days 1-14 (21-day cycles)
+
dexamethasone 20 mg[a] orally on the day of and day after bortezomib
End-of-treatment visit

Treatment until disease progression, death, unacceptable toxicity, withdrawal of consent, or end of study

Primary endpoint: PFS (IRC assessed per IMWG)

Key secondary endpoints: OS, MRD negativity, DOR

Other secondary endpoints: ORR, CRR, ≥ VGPR, TTBR, TTR, TTP, PFS2,[b] AEs, ocular findings, HRQOL, PROs

[a] Patients aged >75 years with comorbidities or intolerance of the 40-mg dose in arm A or 20-mg dose in arm B could have dose level reduced to half per investigator discretion. [b] PFS2 was defined as time from randomization to disease progression after initiation of new antimyeloma therapy or death from any cause, whichever was earliest.

- This post hoc analysis evaluated baseline characteristics and clinical outcomes in LTRs, defined as patients who had PFS ≥30 months
- All patients who have remained on study have been on study for >30 months
 - Data cutoff: July 7, 2025; median follow-up, 36 months in both arms

Post hoc analysis in LTRs and non-LTRs

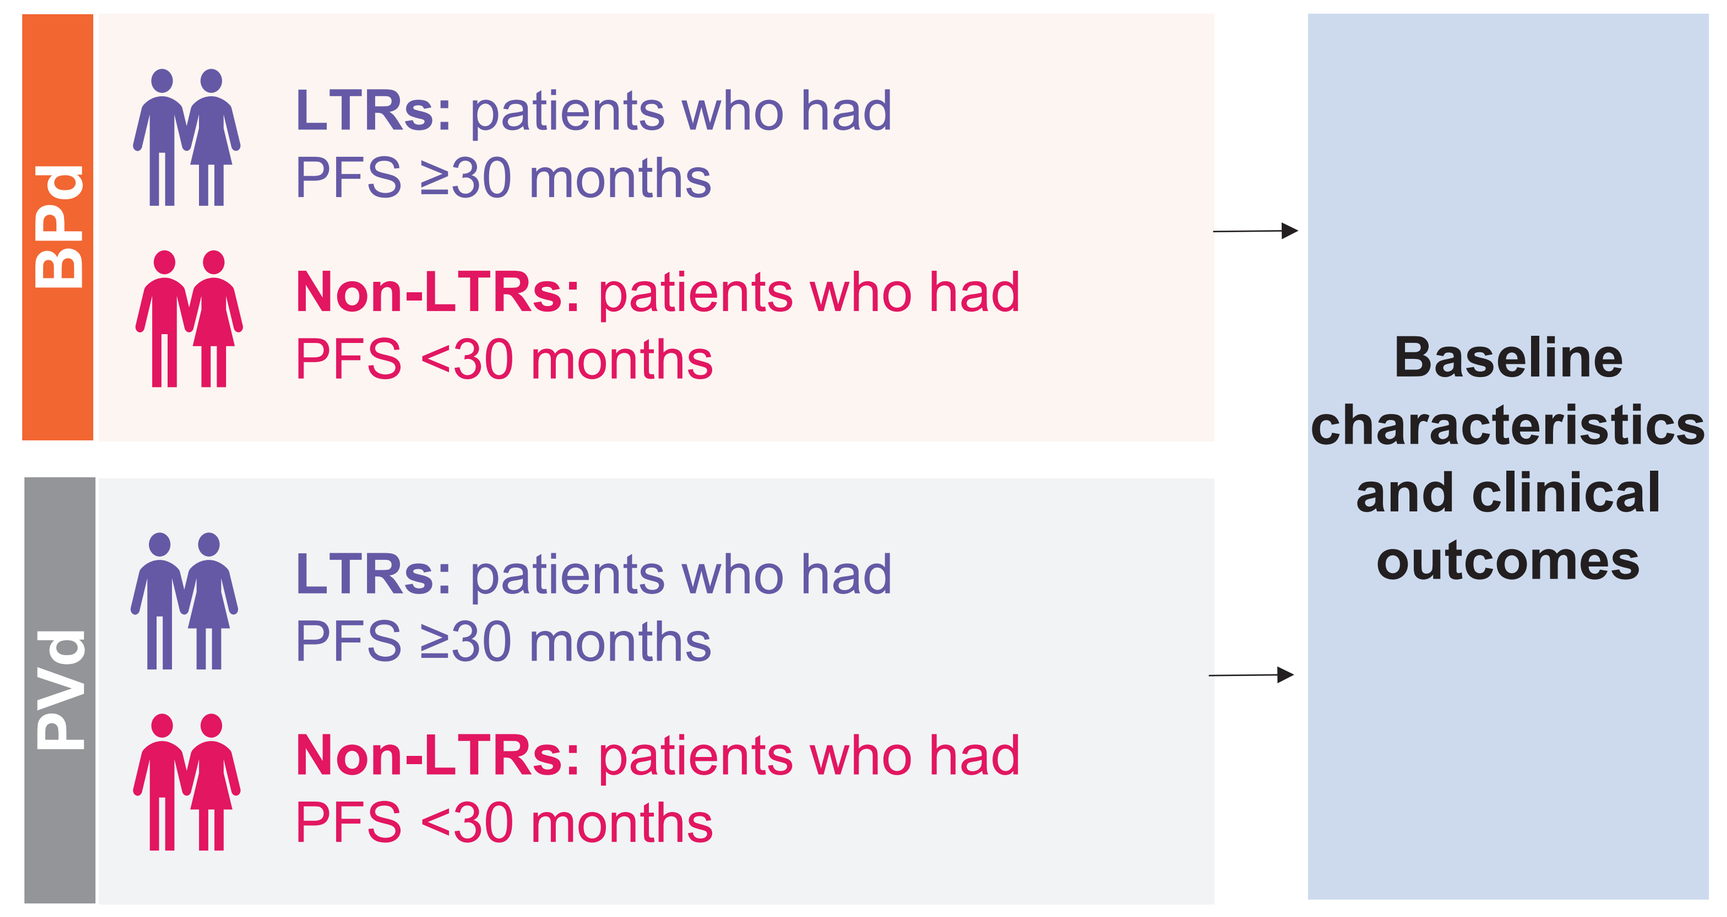

BPd
LTRs: patients who had PFS ≥30 months
Non-LTRs: patients who had PFS <30 months
PVd
LTRs: patients who had PFS ≥30 months
Non-LTRs: patients who had PFS <30 months
Baseline characteristics and clinical outcomes

Results

The proportion of LTRs was higher with BPd, including patients with high-risk cytogenetics

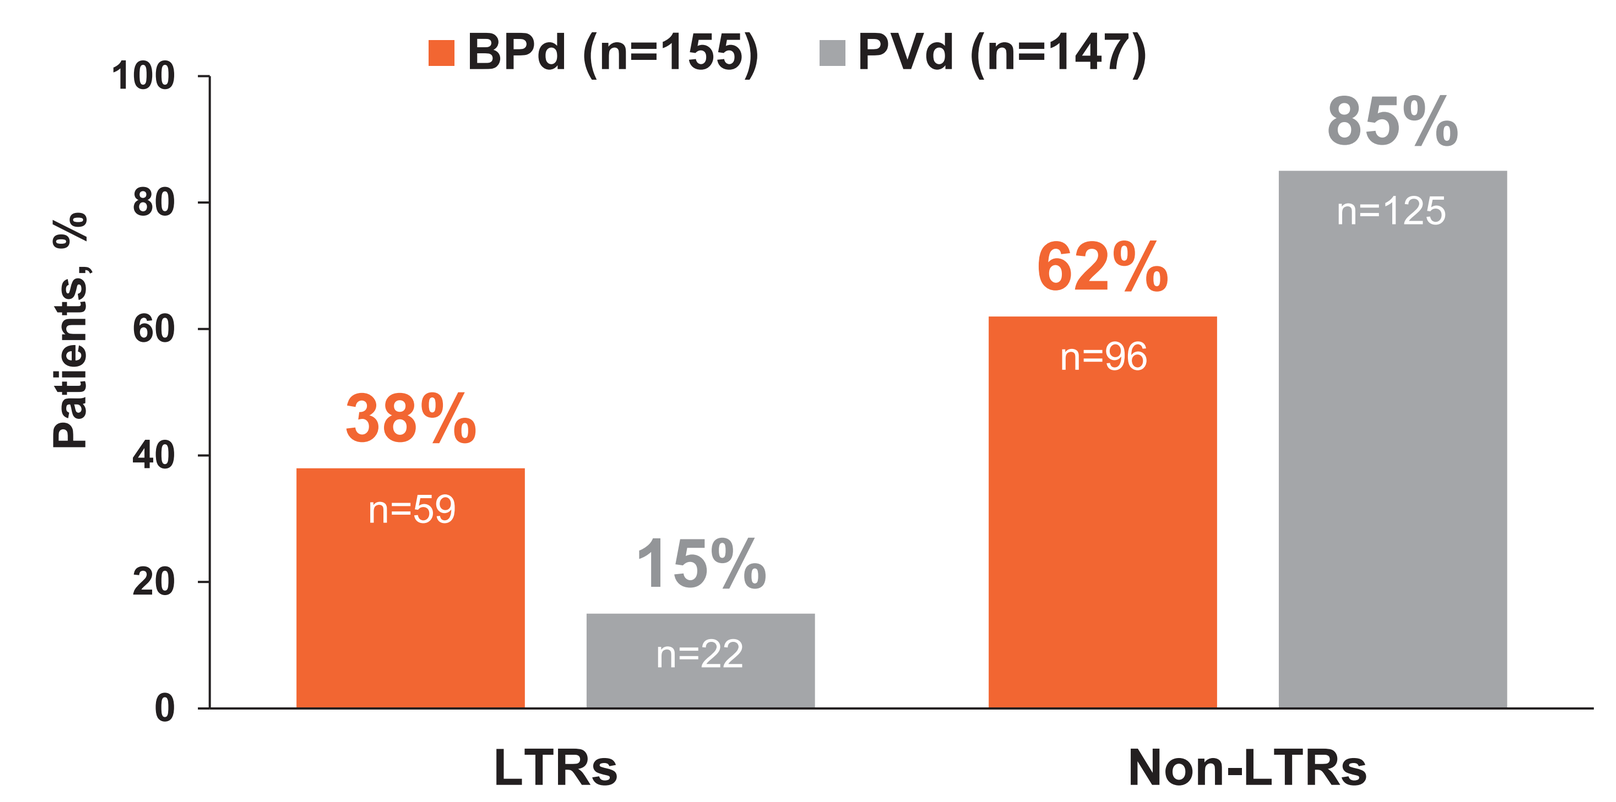

LTRs and Non-LTRs in the BPd and PVd Arms
BPd (n=155)
PVd (n=147)
Patients, %
38%
n=59
15%
n=22
62%
n=96
85%
n=125
LTRs
Non-LTRs

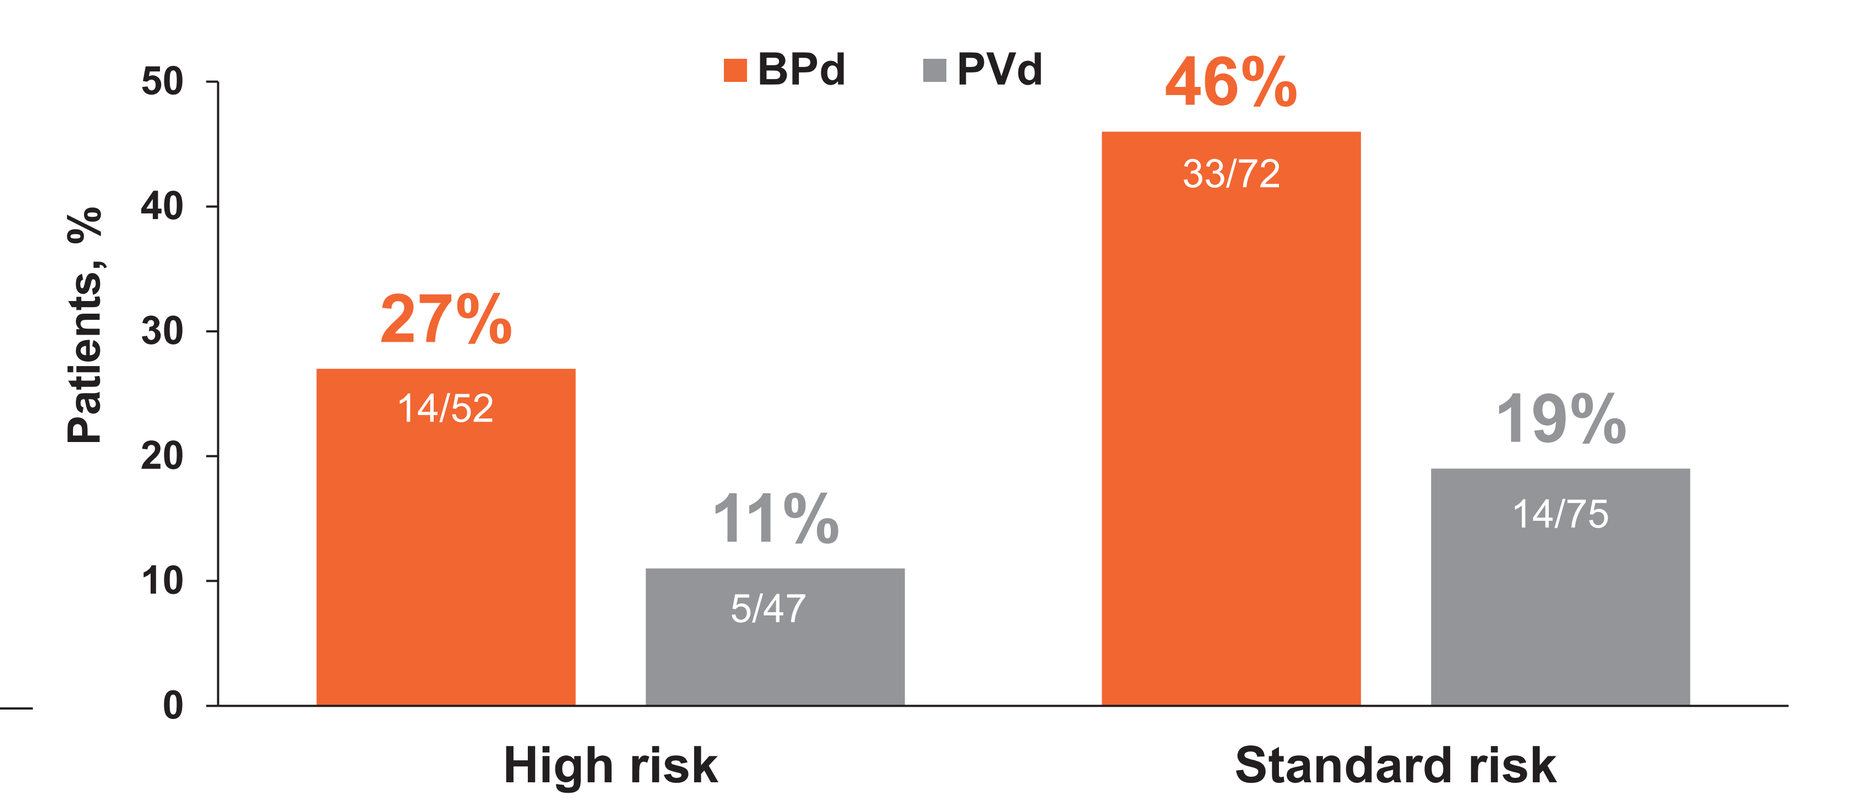

Proportion of LTRs in Patients With High- and Standard-Risk Cytogenetics[a]
BPd
PVd
Patients, %
27%
14/52
11%
5/47
46%
33/72
19%
14/75
High risk
Standard risk

[a] High-risk cytogenetics was defined as the presence of ≥1 of the following: t(4;14), t(14;16), or del(17p13). Standard-risk cytogenetics was defined as negative results for all high-risk abnormalities: t(4;14), t(14;16), and del(17p13). Cytogenetic data were missing or nonevaluable in 12 BPd- and 3 PVd-treated LTRs and 19 BPd- and 22 PVd-treated non-LTRs.

- Of the 302 patients in DREAMM-8, 81 were LTRs

BPd-treated LTRs had deeper responses vs PVd-treated LTRs

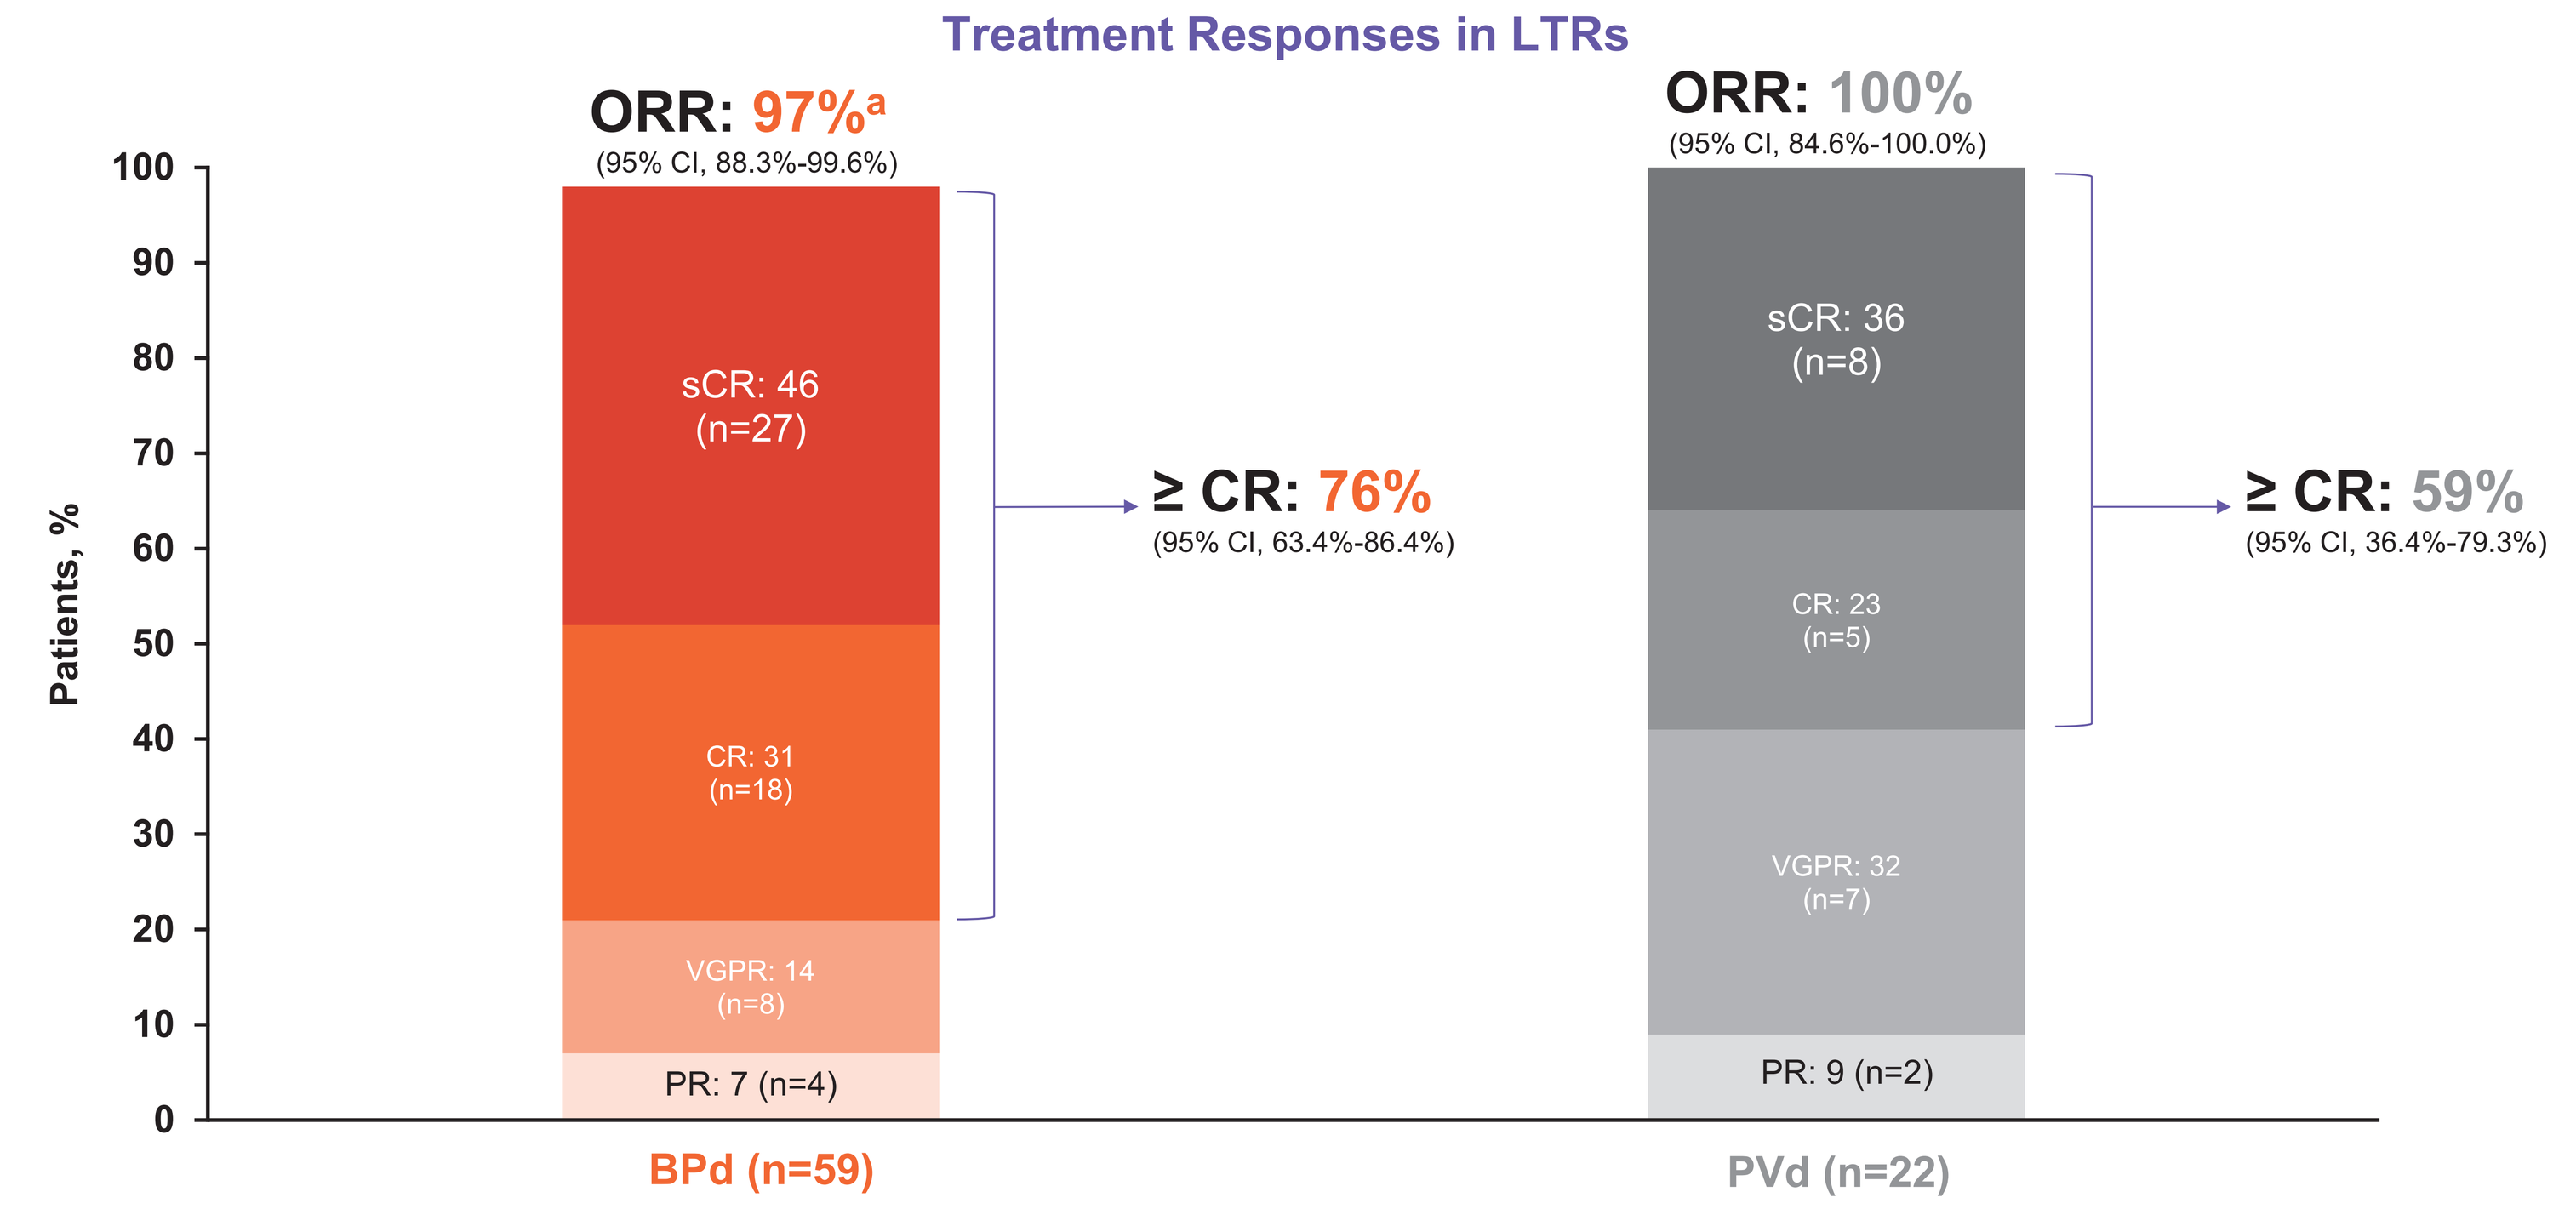

Treatment Responses in LTRs
ORR: 97%[a]
(95% CI, 88.3%-99.6%)
ORR: 100%
(95% CI, 84.6%-100.0%)
Patients, %
sCR: 46 (n=27)
CR: 31 (n=18)
VGPR: 14 (n=8)
PR: 7 (n=4)
≥ CR: 76%
(95% CI, 63.4%-86.4%)
sCR: 36 (n=8)
CR: 23 (n=5)
VGPR: 32 (n=7)
PR: 9 (n=2)
≥ CR: 59%
(95% CI, 36.4%-79.3%)
BPd (n=59)
PVd (n=22)

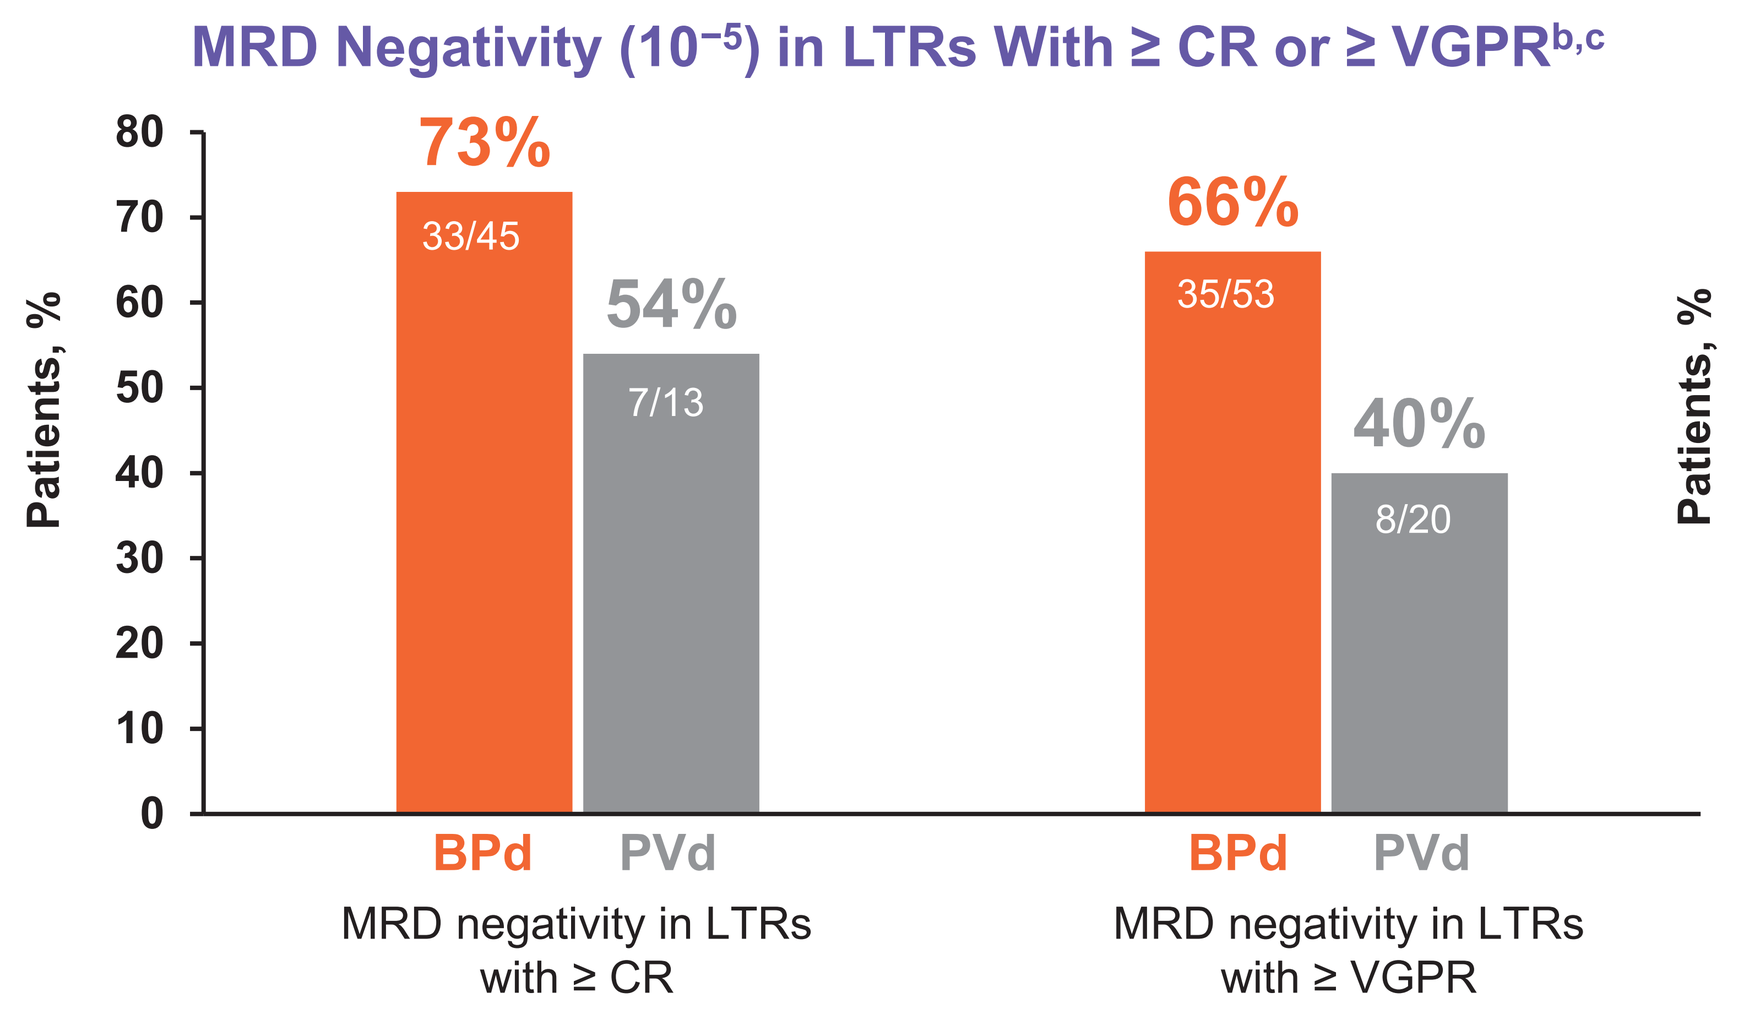

MRD Negativity (10⁻⁵) in LTRs With ≥ CR or ≥ VGPR[b,c]
Patients, %
73%
33/45
54%
7/13
66%
35/53
40%
8/20
BPd PVd
MRD negativity in LTRs with ≥ CR
BPd PVd
MRD negativity in LTRs with ≥ VGPR

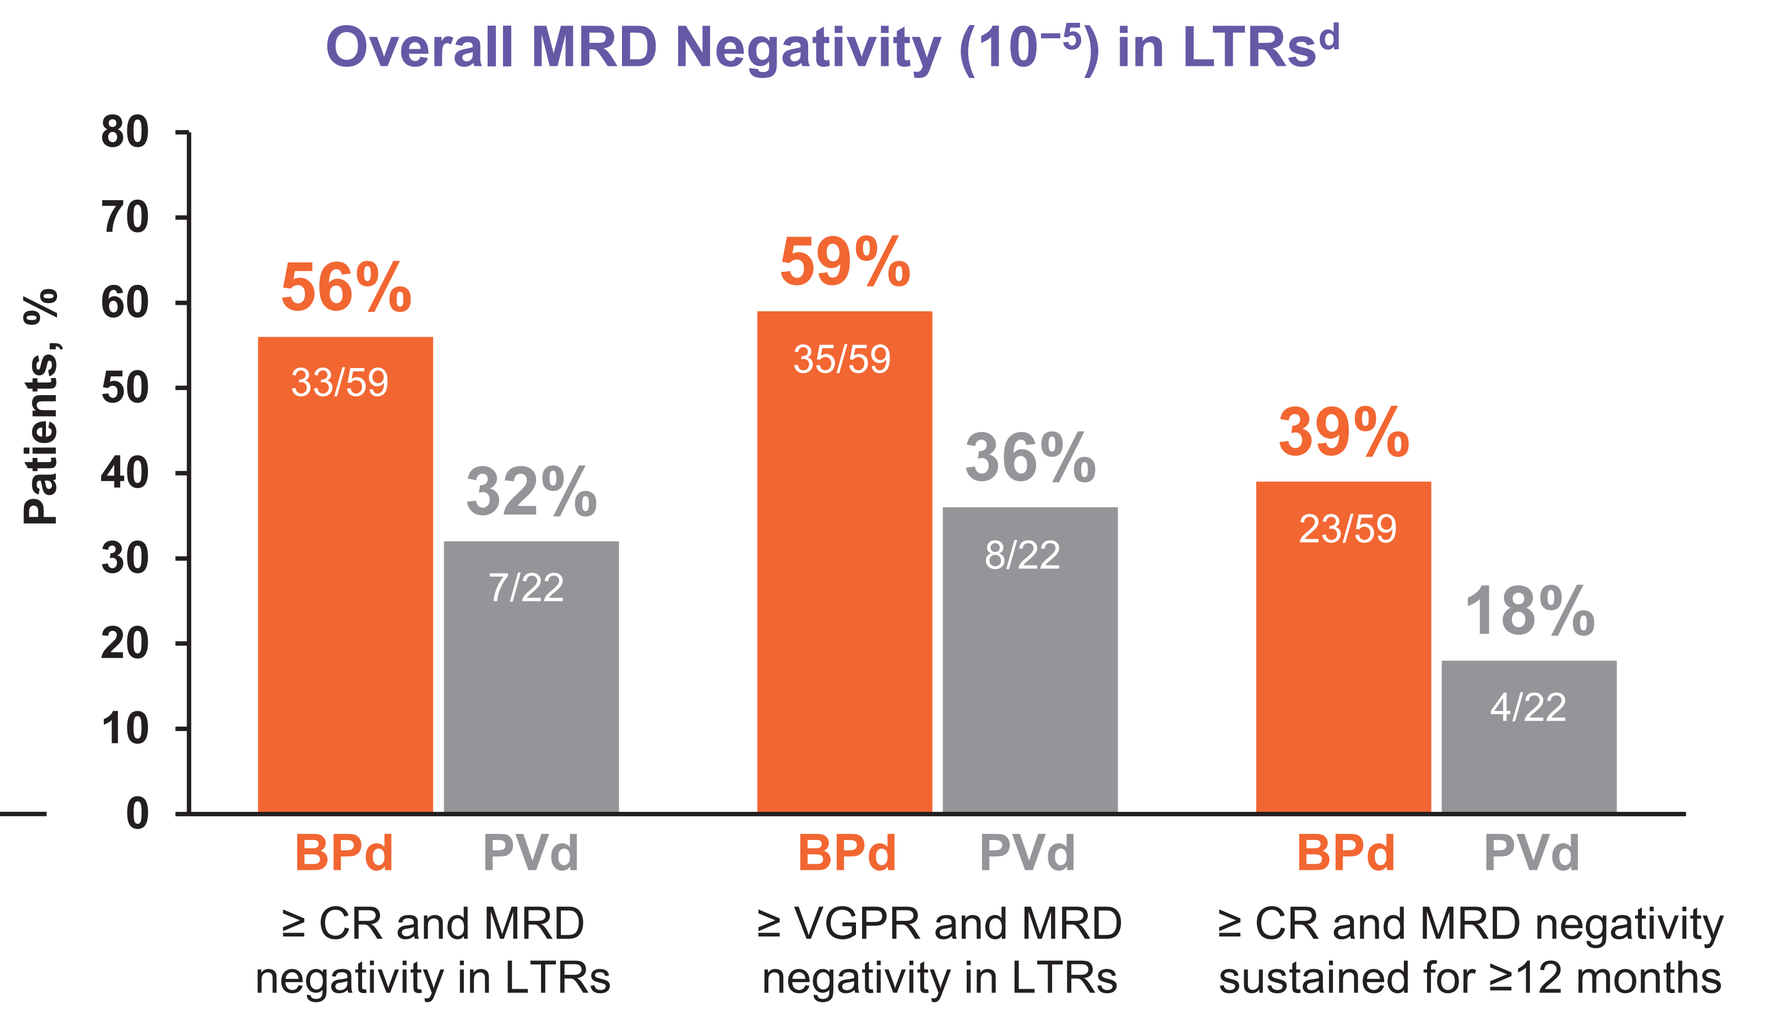

Overall MRD Negativity (10⁻⁵) in LTRs[d]
Patients, %
56%
33/59
32%
7/22
59%
35/59
36%
8/22
39%
23/59
18%
4/22
BPd PVd
≥ CR and MRD negativity in LTRs
BPd PVd
≥ VGPR and MRD negativity in LTRs
BPd PVd
≥ CR and MRD negativity sustained for ≥12 months

[a] ORR was not 100% because 1 patient had minimal response, and 1 patient had stable disease. [b] MRD-negativity rate is defined as the percentage of patients who are MRD negative by next-generation sequencing at a sensitivity of 10^{-5} as assessed by an independent reviewer. [c] MRD-negativity rate calculated based on LTRs with ≥ CR or ≥ VGPR. [d] MRD-negativity rate calculated based on LTRs.

Treatment arms were generally balanced at baseline with a higher proportion of LTRs in earlier therapy lines

	LTRs	
Baseline characteristics	BPd (n=59)	PVd (n=22)
Female/male sex, n (%)	25 (42) / 34 (58)	12 (55) / 10 (45)
Age, median (range), years	65.0 (44-79)	69.5 (42-79)
ISS at screening, n (%)		
I	39 (66)	15 (68)
II	15 (25)	6 (27)
III	5 (8)	1 (5)
Line of therapy completed at screening, n (%)		
1	39 (66)	15 (68)
2	14 (24)	4 (18)
≥3	6 (10)	3 (14)

- The safety profile in LTRs was generally consistent with previous reports in the overall DREAMM-8 safety population[3]
- In BPd vs PVd LTRs, exposure-adjusted rates per 100 patient-years of neutropenia[a] (23.4 vs 21.1), infections[b] (27.4 vs 27.6), and pneumonia (9.6 vs 9.2) were similar, as were exposure-adjusted rates of grade 3/4 events (30 vs 25)
- Grade ≥3 exposure-adjusted rates per 100-patient years of infections[b] (17.8 vs 6.6) and pneumonia (8.1 vs 3.9) were higher for BPd vs PVd LTRs

[a] Neutropenia includes preferred terms febrile neutropenia, neutropenia, and neutrophil count decreased. [b] System organ class event.

BPd-treated LTRs demonstrated prolonged PFS that continued following first subsequent antimyeloma therapy

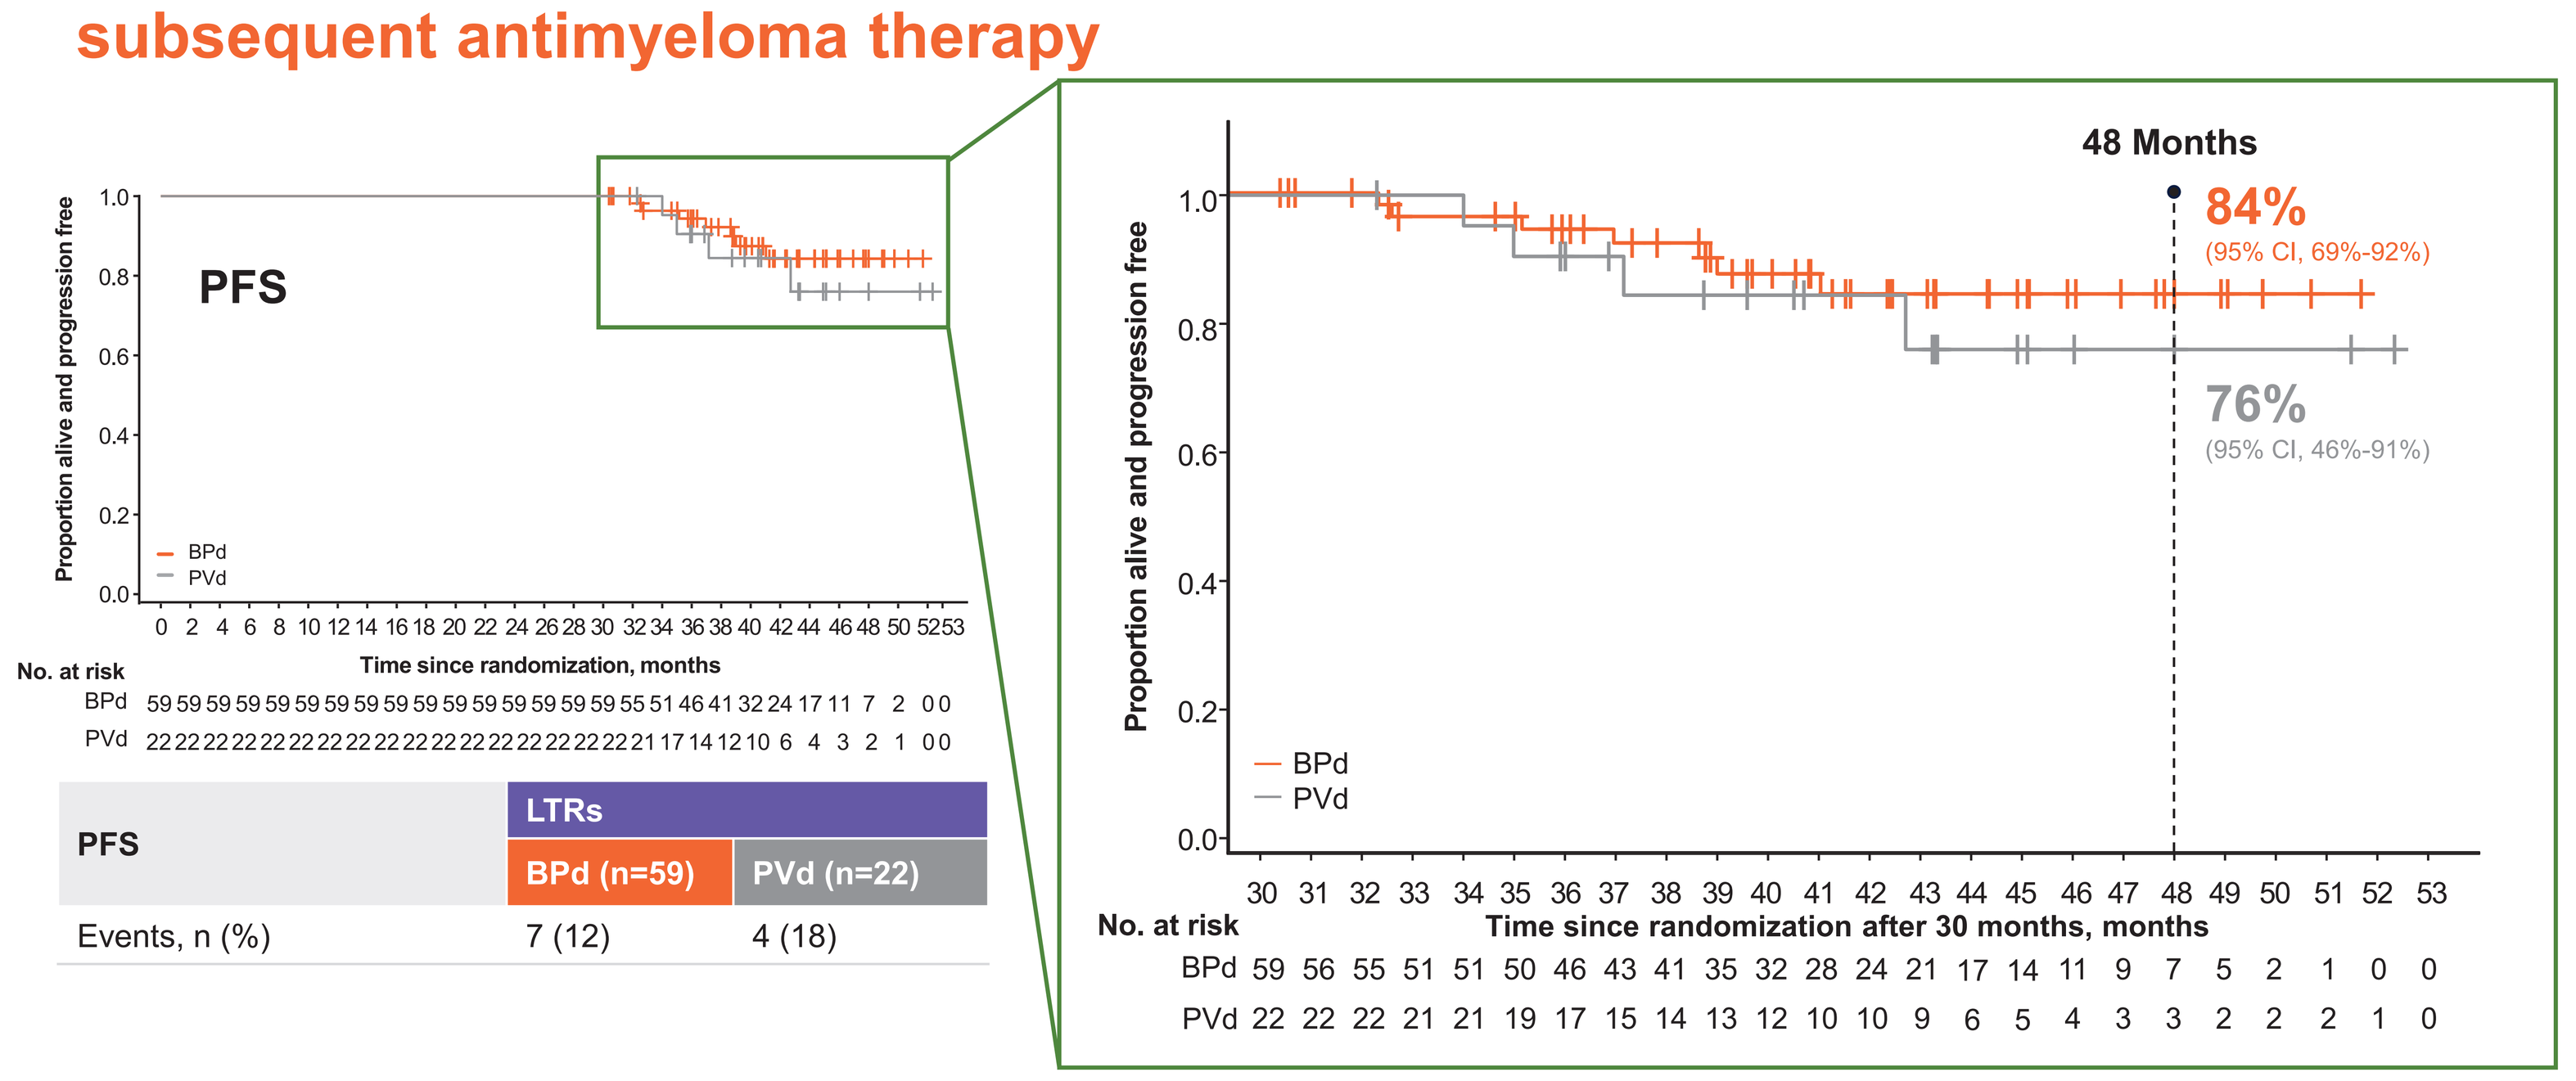

PFS
Proportion alive and progression free
BPd
PVd
Time since randomization, months
No. at risk
BPd 59 59 59 59 59 59 59 59 59 59 59 59 59 59 59 59 55 51 46 41 32 24 17 11 7 2 0 0
PVd 22 22 22 22 22 22 22 22 22 22 22 22 22 22 22 22 21 17 14 12 10 6 4 3 2 1 0 0
48 Months
84%
(95% CI, 69%-92%)
76%
(95% CI, 46%-91%)
Time since randomization after 30 months, months
No. at risk
BPd 59 56 55 51 51 50 46 43 41 35 32 28 24 21 17 14 11 9 7 5 2 1 0 0
PVd 22 22 22 21 21 19 17 15 14 13 12 10 10 9 6 5 4 3 3 2 2 2 1 0

PFS	LTRs	
	BPd (n=59)	PVd (n=22)
Events, n (%)	7 (12)	4 (18)

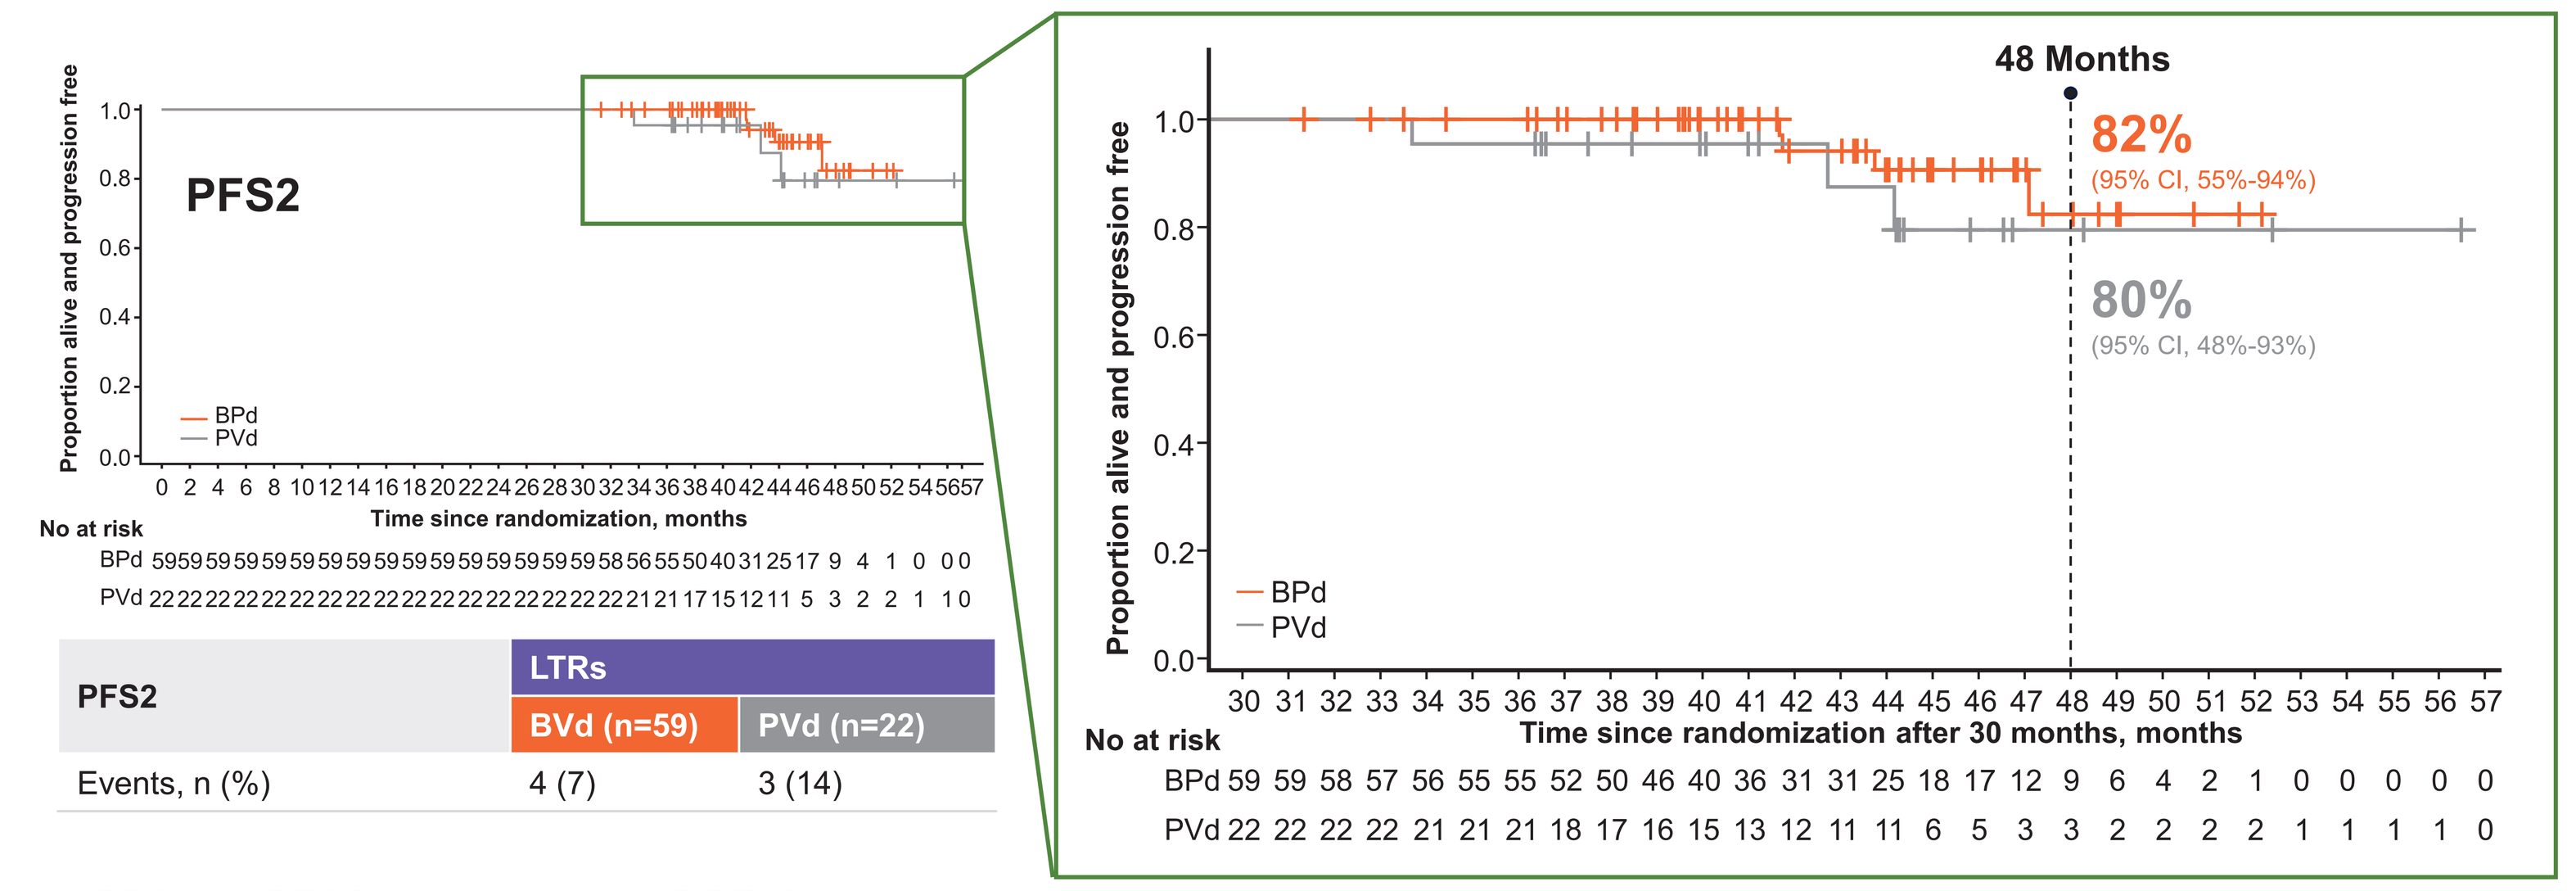

PFS2
Proportion alive and progression free
BPd
PVd
Time since randomization, months
No at risk
BPd 59 59 59 59 59 59 59 59 59 59 59 59 59 59 59 59 58 56 55 50 40 31 25 17 9 4 1 0 0 0
PVd 22 22 22 22 22 22 22 22 22 22 22 22 22 22 22 22 21 21 17 15 12 11 5 3 2 2 1 1 0
48 Months
82%
(95% CI, 55%-94%)
80%
(95% CI, 48%-93%)
Time since randomization after 30 months, months
No at risk
BPd 59 59 58 57 56 55 55 52 50 46 40 36 31 31 25 18 17 12 9 6 4 2 1 0 0 0 0 0
PVd 22 22 22 22 21 21 21 18 17 16 15 13 12 11 11 6 5 3 3 2 2 2 2 1 1 1 1 0

PFS2	LTRs	
	BVd (n=59)	PVd (n=22)
Events, n (%)	4 (7)	3 (14)

- PFS and PFS2 follow-up in the DREAMM-8 study is ongoing

Abbreviations

ADC, antibody-drug conjugate; AE, adverse event; BCMA, B-cell maturation antigen; belamaf, belantamab mafodotin; BPd, belamaf, pomalidomide, and dexamethasone; CR, complete response; CRR, complete response rate; DOR, duration of response; HR, hazard ratio; HRQOL, health-related quality of life; IMWG, International Myeloma Working Group; IRC, independent review committee; ISS, International Staging System; IV, intravenous; LOT, line of therapy; LTR, long-term responder; MRD, minimal residual disease; ORR, overall response rate; OS, overall survival; Pd, pomalidomide and dexamethasone; PFS, progression-free survival; PFS2, progression-free survival on subsequent line of therapy; PR, partial response; PRO, patient-reported outcome; PVd, pomalidomide, bortezomib, and dexamethasone; Q4W, every 4 weeks; RRMM, relapsed/refractory multiple myeloma; SC, subcutaneous; sCR, stringent complete response; TTBR, time to best response; TTP, time to progression; TTR, time to response; VGPR, very good partial response.

References

1. Amin R, et al. *Med Oncol.* 2025;42:204.
2. Chan CH, et al. *Mol Ther Oncol.* 2025;33(4):201044.
3. Dimopoulos MA, et al. *N Engl J Med.* 2024;391(5):408-421.

Acknowledgments

The DREAMM-8 trial (NCT04484623) was sponsored by GSK (study ID: 207499).

We thank the patients who volunteered to participate in the trial, along with their families and caregivers, and the physicians and nurses who cared for the patients and supported this clinical trial.

Drug-linker technology was licensed from Seagen Inc; monoclonal antibody was produced using POTELLIGENT Technology, licensed from BioWa.

Medical writing and editorial assistance was provided by Nucleus Global, an Inizio company, and funded by GSK.

Disclosures

MAD reports payment or honoraria for lectures, presentations, speakers bureaus, manuscript writing, or educational events from Amgen, Sanofi, Regeneron, Menarini, Takeda, GSK, BMS, Janssen, BeiGene, Swixx, and AstraZeneca. **MB** reports payment or honoraria from BMS and Johnson & Johnson and a consulting role for GSK, Takeda, Amgen, Janssen, and Menarini. **SD** reports payment or honoraria from Amgen, GSK, Janssen, and Takeda. **VV** reports honoraria from AbbVie, Amgen, AstraZeneca, BeiGene, Gilead Sciences, Janssen, Novartis, Roche, Sanofi, Takeda, BioCad, VMSi, and MSD; a consulting role for AbbVie, AstraZeneca, BeiGene, Gilead Sciences, Janssen Oncology, Novartis, Roche, Sanofi, and Takeda; and support for meeting attendance and/or travel from AstraZeneca. **HQ** reports research support from GSK, BMS, AbbVie, Antengene, and Karyopharm; payment or honoraria from BMS, Johnson & Johnson, GSK, and AbbVie; expert testimony for Pfizer; and a consulting role for BMS, Johnson & Johnson, GSK, Regeneron, and Pfizer. **IŠ** reports consulting fees from BMS, Amgen, Janssen-Cilag, Takeda, Sanofi, and GSK; honoraria from Amgen, Janssen-Cilag, Takeda, BMS, and Sanofi; support for meeting attendance and/or travel from Janssen-Cilag, BMS, and Sanofi; and participation on data safety monitoring or advisory boards with Janssen-Cilag, Takeda, Sanofi, and GSK. **JR** reports consulting fees from GSK, Johnson & Johnson, and Sanofi; honoraria from Johnson & Johnson, Pfizer, BMS, Sanofi, and GSK; support for meeting attendance and/or travel from Johnson & Johnson and Sanofi; and participation on data safety monitoring or advisory boards with GSK, Johnson & Johnson, Sanofi, BMS, and Pfizer. **PR** reports no disclosures. **KK** reports research support from Sanofi, Johnson & Johnson, and BMS and consulting fees from Johnson & Johnson, BMS, AbbVie, and GSK. **SZ** reports no disclosures. **AM**, **KM**, **MP**, **KB**, **JG**, and **EEM** report employment and stock or stock options from GSK. **M-VM** reports honoraria from Janssen, BMS, Amgen, GSK,Sanofi, AbbVie, Kite, Stemline, and Pfizer and participation on data safety monitoring or advisory boards with Janssen, BMS, Amgen, Sanofi, GSK, Roche, Pfizer, AbbVie, Kite, and Stemline. **ST** reports honoraria from Amgen, Forus, GSK, Janssen, Pfizer, and Sanofi; a consulting role for Bristol Myers Squibb, GSK, and Roche; and receipt of research support from Amgen, Bristol Myers Squibb, Genentech, GSK, Janssen, K36 Therapeutics, Pfizer, and Roche.

Society of Hematologic Oncology (SOHO) 2026 Annual Meeting | September 9-12, 2026 | Houston, TX, USA

© 2026 American Society of Clinical Oncology (ASCO), Inc. Reused with permission. This poster was previously presented at the 2026 ASCO Annual Meeting. All rights reserved.

Presenting Author: Meletios A. Dimopoulos, mdimop@med.uoa.gr

GSK