IBCL-1332

Phase 3 Results From the EPCORE FL-1 Trial of Epcoritamab With Lenalidomide and Rituximab (R^2) Versus R^2 for Relapsed or Refractory Follicular Lymphoma

Bradley D. Hunter,* Lorenzo Falchi,[1] Marcel Nijland,[2] Huiqiang Huang,[3] Kim M. Linton,[4] John F. Seymour,[5] Rong Tao,[6] Michal Kwiatek,[7] Abel Costa,[8] Theodoros P. Vassilakopoulos,[9] Richard Greil,[10] Ana Jiménez-Ubieto,[11] Shane Gangatharan,[12] Ohad Benjamini,[13] Catherine Thieblemont,[14] Alessandra Tucci,[15] Anna Elinder-Camburn,[16] Arpad Illes,[17] Jan Novak,[18] Miguel Pavlovsky,[19] Andrew McDonald,[20] Dok Hyun Yoon,[21] Yuko Mishima,[22] Gauri Sunkersett,[23] JP Mei,[23] Nabanita Mukherjee,[23] Feng Zhu,[23] Elena Favaro,[24] Franck Morschhauser[25]

*Presenting on behalf of the EPCORE FL-1 study investigators, Intermountain LDS Hospital, Salt Lake City, Utah, USA; [1]Lymphoma Service, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]University Medical Center Groningen, University of Groningen, Groningen, Netherlands; [3]Sun Yat-sen University, Guangzhou, China; [4]The Christie NHS Foundation Trust, Manchester Cancer Research Centre, and Division of Cancer Sciences, University of Manchester, Manchester, UK; [5]Peter MacCallum Cancer Centre and The Royal Melbourne Hospital, Melbourne, Australia; [6]Fudan University Cancer Hospital, Shanghai, China; [7]Aidport Clinical Trials Hospital, Skorzewo (Poznan), Poland; [8]Instituto D'Or de Pesquisa e Ensino, São Paulo, Brazil; [9]Laikon General Hospital, National and Kapodistrian University of Athens, Athens, Greece; [10]Salzburg Cancer Research Institute-Center for Clinical Cancer and Immunology Trials (SCRICCCIT); Individualized Immunotherapy Research and Development (iiT) GmbH; Salzburg Cancer Research Institute-Laboratory of Immunological and Molecular Cancer Research, Salzburg, Austria; [11]Hospital Universitario 12 de Octubre, Madrid, Spain; [12]Fiona Stanley Hospital, Murdoch, Australia; [13]The Chaim Sheba Medical Center, Ramat Gan, Israel; [14]Hôpital Saint-Louis, Paris, France; [15]ASST degli Spedali Civili di Brescia, Brescia, Italy; [16]North Shore Hospital, Auckland, New Zealand; [17]Debreceni Egyetem-Klinikai Kozpont, Debrecen, Hungary; [18]Fakultni nemocnice Kralovske Vinohrady, Prague, Czechia; [19]FUNDALEU, Buenos Aires, Argentina; [20]Alberts Cellular Therapy, Gauteng, South Africa; [21]Asan Medical Center, University of Ulsan, College of Medicine, Seoul, South Korea; [22]Cancer Institute Hospital, Japanese Foundation for Cancer Research, Tokyo, Japan; [23]AbbVie, North Chicago, IL, USA; [24]Genmab, Copenhagen, Denmark; [25]Hôpital Claude Huriez, Lille, France

Objective

- We present pivotal efficacy and safety data from the phase 3 EPCORE FL-1 trial (NCT05409066) of epcoritamab+R^2 versus R^2 alone for R/R FL

Conclusion

- In this phase 3 trial in patients with 2L+ FL, fixed-duration epcoritamab plus R^2 was superior to R^2
 - 79% reduction in the risk of disease progression or death
 - Higher rates and durable deep responses (CRR: 83% for epcoritamab+R^2, 50% for R^2; DOCR: not reached for epcoritamab+R^2, 18.6 months for R^2)
 - Benefit was consistent across broad patient subgroups, including both low- and high-risk populations
 - Superior TTNLT and positive trend in OS for epcoritamab+R^2
- The safety profile was manageable in the outpatient setting and remained consistent with the known toxicities of the individual components
- Grade ≥ 3 infection risk peaked in cycles 1 and 2 and stabilized from cycle 4 onward
- Epcoritamab+R^2 is a novel chemotherapy-free, fixed-duration therapy suitable for outpatient administration, and this regimen sets a new benchmark as standard of care in 2L+ FL

Copies of this poster obtained through quick response (QR) code are for personal use only and may not be reproduced without permission from the authors of this poster.

Acknowledgments

We thank the patients, their families and caregivers, study investigators, and site personnel for their participation in this study.

This study was funded by AbbVie and Genmab A/S. All authors had access to relevant data and participated in the drafting, review, and approval of this presentation. No honoraria or payments were made for authorship. We also wish to thank Rastislav Moskal, PharmD, of AbbVie for scientific contributions, Alena Vajikova, MS, of AbbVie for study management, Yukun Li, PhD, of AbbVie for statistical analyses, and Ruiling Zhang, MS, of AbbVie for statistical programming. Medical writing support was provided by Kristina Y. Aguilera, PhD, and funded by AbbVie. Medical writing support for this encore presentation was provided by Laura Ruhge, PhD, of Bio Connections LLC, and funded by AbbVie.

References

1. Rivas-Delgado A, et al. *Br J Haematol*. 2019;184(5):753–9.
2. Leonard JP, et al. *J Clin Oncol*. 2019;37(14):JCO.19.00010.
3. FDA. EPKINLY (Epcoritamab) [Prescribing Information] Plainsboro, NJ. 2025.
4. EMA. Tepkinly: Annex I Summary of Product Characteristics.
5. Epkinly [Japan package insert]. AbbVie GK, Genmab KK. 2025.
6. Engelberts PJ, et al. *EBioMedicine*. 2020;52:102625.
7. Golay J, et al. *Antibodies*. 2020; 9(4):58.
8. Gribben JG, et al. *J Clin Oncol*. 2015;33(25):2803–11.
9. Dandamudi DB, et al. *Leuk Lymphoma* 2025; 66: 1688–99.
10. Falchi L, et al. *Blood*. 2024;144 (Supplement 1):342.
11. Vose J, et al. *J Clin Oncol*. 2024;42(16_suppl):7015–7015.

Disclosures

Financial arrangements of the authors with companies whose products may be related to the present report are listed as declared by the authors: **BDH** has served in a consultancy role and received honoraria from Kite, Novartis, BMS, Poseida Therapeutics, Caribou Biosciences, and Kyverna; and has received research funding and honoraria from Johnson & Johnson. **LF** has received research funding from Roche, Genentech, Genmab, AbbVie, Innate Pharma, BeOne, AstraZeneca; consultancy fees from Roche, Genentech, Genmab, AbbVie, Sanofi, AstraZeneca, Merck; served in an advisory role for AbbVie, Genentech, ADC Therapeutics, Seagen, Ipsen, Johnson & Johnson, Regeneron; and received honoraria from Roche, Genmab, AbbVie, Kite. **MN** received research funding from Takeda and consulting fees from AbbVie. All payments to institution. **KML** served in a consultancy role for Kite, Gilead, Genmab, Roche, AbbVie, BeiGene, Bristol Myers Squibb, and Celgene; received research funding from Kite/Gilead, Genmab, Roche, AbbVie, BeiGene, Bristol Myers Squibb, ADC Therapeutics, AstraZeneca, CellCentric, Janssen, MorphoSys, MSD, Nurix, Regeneron, Step Pharma, Viracta, and Celgene; participated on speakers bureaus for AbbVie, Bristol Myers Squibb, and Celgene; received travel support from Bristol Myers Squibb and Celgene; has served as a member of the Epcoritamab Global Council; and received research funding from the National Institute for NIHR Manchester Biomedical Research Center (NIHR203308). **JFS** served in a consultancy role for TG Therapeutics and Bristol Myers Squibb; received research funding from AbbVie, Bristol Myers Squibb, Janssen, Loxo and Roche; and received honoraria from AbbVie, AstraZeneca, BeiGene, Bristol Myers Squibb, Gilead, Janssen, Loxo and Roche. **RT** has served in a consultancy role for AbbVie. **AC** has received honoraria from J&J, AbbVie, AstraZeneca, Lilly, Knight, BeiGene, Regeneron, IPSEN and Pfizer and research funding from AbbVie. **TPV** has received research support/PI for Merck, Roche, AbbVie, Bristol Myers Squibb and Karyopharm; received honoraria from Takeda, Roche, Genesis Pharma, AbbVie, Gilead, Sandoz, AstraZeneca, Lilly, Janssen, Sobi and Swixx; and served in an advisory role for Roche, Genesis Pharma, Takeda, AbbVie, Merck, Gilead, Sandoz, Teva, AstraZeneca, Winmedica, Sobi, Swixx and Lilly. **RG** received research grants from Celgene, Roche, Merck, Takeda, AstraZeneca, Novartis, Amgen, Bristol Myers Squibb, MSD, Sandoz, AbbVie, Gilead and Daiichi Sankyo; received consulting fees or other remuneration (payment) from Celgene, Novartis, Roche, Bristol Myers Squibb, Takeda, AbbVie, AstraZeneca, Janssen, MSD, Amgen, Merck, Gilead, Daiichi Sankyo and Sanofi; held stock from Novo Nordisk and Lilly; and has other conflicts of interests with Roche, Amgen, Janssen, Astra Zeneca, Novartis, MSD, Celgene, Gilead, Bristol Myers Squibb, and AbbVie. **AJU** reports consultancy/advisory role for Regeneron, Kite/Gilead, Lilly, AbbVie, AstraZeneca and Incyte; served on speakers bureaus for Janssen, Sandoz, Roche, Incyte, Kite/Gilead and AbbVie; and served in advisory boards for AbbVie, AstraZeneca, Kite/Gilead, Incyte, Lilly, and Regeneron. **OB** has served in a consulting role for AbbVie, Janssen, Roche, AstraZeneca and Lilly; received research funding and speaker fees from AbbVie; and served in an advisory role for Lilly and AbbVie. **CT** received honoraria from ADC Therapeutics, AstraZeneca, Incyte, Sanofi, Amgen, Novartis, Cellectis, Janssen, Bayer, Roche, BeiGene, AbbVie, Bristol Myers Squibb/Celgene, Takeda, Kite/Gilead and Regeneron; served in a consultancy role for Novartis, Janssen, BeiGene, AbbVie, Bristol Myers Squibb/Celgene, Takeda, Kite/Gilead and Regeneron; received research funding from Roche, AbbVie and Kite/Gilead; and was previously employed by the University of Paris. **AT** has received honoraria from Takeda, Regeneron, Novartis, AbbVie, Gilead-Kite, Incyte, AstraZeneca, Lilly, Kiowa Kyrin and BeiGene. **AE-C** reports serving in an advisory role and receiving speaker fees from AbbVie and Janssen and receiving travel support from Roche. **AI** served in a consultancy role for AbbVie, Bristol Myers Squibb and Novartis and received honoraria from AstraZeneca and Takeda. **MP** reports serving in an advisory role and as a speaker for Janssen, AbbVie and AstraZeneca; participating on an advisory board for AbbVie, AstraZeneca, and Janssen; and receiving travel grants from Sanofi, Amgen and Roche. **AM** reports research funding from AbbVie, Roche, MSD and BeOne and receiving honoraria from BeOne, Biomab Innovation and Roche. **DHY** reports serving in a consultancy role for AbbVie, Abclon, BeOne, Bristol Myers Squib, GI cell, GC cell, Verismo, J&J, Novartis, Regeneron and Roche; receiving patents & royalties from Boryung; receiving research funding from AbbVie, BeOne, Boryung, Celltrion, Kyowa Kirin, Janssen and Samyang; and receiving honoraria from AbbVie, Bristol Myers Squib, Boryung, GSK, Kyowa Kirin, Novartis, Roche, Takeda and J&J. **GS**, **JPM**, **NM**, and **FZ** report employment by AbbVie and own AbbVie stock or options. **EF** reports employment by Genmab and owns Genmab stock or options. **FM** reports serving in a consultancy role for Kite/Gilead, Roche, AbbVie, Bristol Myers Squibb, Janssen, Takeda and Miltenyi; serving on speakers bureaus for Roche and Kite/Gilead; receiving honoraria from AstraZeneca; and participation on an advisory board for AbbVie, Bristol Myers Squibb, Janssen, Kite/Gilead, Miltenyi, Roche and Takeda. **HH**, **MK**, **SG**, **JN**, and **YM** declare no competing interests.

Presented at the Annual Meeting of the Society of Hematologic Oncology; September 9–12, 2026; Houston, Texas; DV-021258

Introduction

- Most patients with follicular lymphoma (FL) experience disease relapse and the duration of remission is shorter with each subsequent line of therapy,[1] underscoring the need for more effective therapies in earlier lines
- The combination of lenalidomide plus rituximab (R^2) is a chemotherapy-free standard of care in relapsed/refractory (R/R) FL; however, outcomes remain suboptimal for many patients[2]
- Epcoritamab is a CD3×CD20 bispecific antibody, approved as monotherapy for R/R FL after ≥ 2 prior lines of treatment,[3–5] which when combined with lenalidomide and rituximab may augment antilymphoma activity and lead to improved clinical outcomes in patients with R/R FL[6–8]
 - Unique among CD3×CD20 T-cell engagers, preclinical studies have demonstrated minimal interference between epcoritamab and rituximab antitumor activity when combined[9]
- Early data from the phase 1b/2 EPCORE® NHL-2 trial (NCT04663347) with fixed-duration epcoritamab plus R^2 for R/R FL showed deep and durable responses with encouraging clinical outcomes and manageable safety[10]

Study Design

EPCORE FL-1: Phase 3, Global, Randomized, Open-Label Study

Key eligibility criteria
- Histologically confirmed CD20+ FL
- Grade 1-3a, Stage II-IV
- ≥ 1 prior treatment including anti-CD20 mAb plus an alkylating agent
- Met ≥ 1 GELF criterion

Stratification factors
- Disease status:
 - 2L: > or ≤ 2 years since last therapy
 - 3L+: ≥ or < 6 months since last therapy
- Region: US/EU vs Rest of World

Randomization 1:1

Fixed-Duration: 12 Cycles (28-Day Cycles)

Epcoritamab (48 mg) plus R^2
- **Epcoritamab** (3-SUD cycle 1: QW;[a,b] cycles 2–3, QW; cycles 4–12, Q4W)
- **Rituximab** (375 mg/m^2), 5 cycles (cycle 1, QW; cycles 2–5, Q4W)
- **Lenalidomide** (20 mg), 12 cycles (cycle 1–12, QD, D1–21)

R^2
- **Rituximab** (375 mg/m^2), 5 cycles (cycle 1, QW; cycles 2–5, Q4W)
- **Lenalidomide** (20 mg), 12 cycles (cycle 1–12, QD, D1–21)

- **Dual primary endpoints: ORR per IRC and PFS per IRC**
- Key secondary endpoints: CR rate per IRC, OS, and MRD[c]
- Additional secondary endpoints: DOR, DOCR, TTNLT, safety, and PRO assessments

Data cutoff: May 24, 2025; Median follow-up: 14.8 months[d]
Enrollment period: October 2022 – January 2025

[a]Two step-up dosing (SUD) regimens during cycle 1 to mitigate the risk of cytokine release syndrome: either a 2-SUD (0.16 mg on cycle 1 day 1, 0.8 mg on cycle 1 day 8), or 3-SUD (0.16 mg on cycle 1 day 1, 0.8 mg on cycle 1 day 8, 3 mg on cycle 1 day 15) regimen, followed by full dose 48 mg. The 3-SUD regimen was implemented after reduced CRS severity and incidence had been observed in the EPCORE NHL-1 FL trial (NCT03625037).[11] [b]The 24 mg epcoritamab plus R^2 arm was closed to enrollment based on the superior efficacy for the 48 mg dose from EPCORE NHL-2.[10] Only the data for the optimal dose explored (48 mg) are presented here. [c]Minimal residual disease data are forthcoming in a future analysis. [d]The data presented here are from the second planned interim analysis (May 24, 2025) after 78% Information Fraction for PFS had occurred.

Results

Table 1: Baseline Demographics and Disease Characteristics Were Generally Balanced

Characteristic	Epcoritamab+R^2 (N = 243)	R^2 (N = 245)	Overall (N = 488)
Median age, years (range)	60 (30, 84)	63 (24, 89)	61 (24, 89)
≥ 65, n (%)	88 (36)	106 (43)	194 (40)
Male, n (%)	139 (57)	138 (56)	277 (57)
Race, n (%)			
Asian	63 (26)	54 (22)	117 (24)
Black	6 (2)	2 (< 1)	8 (2)
White	168 (69)	184 (75)	352 (72)
Ethnicity, n (%)			
Hispanic	29 (12)	28 (11)	57 (12)
ECOG, n (%)			
0	166 (68)	170 (69)	336 (69)
1–2	77 (32)	75 (31)	152 (31)
Ann Arbor stage, n (%)			
II	37 (15)	44 (18)	81 (17)
III–IV	206 (85)	201 (82)	407 (83)
FLIPI score, n (%)			
0–1	63 (26)	56 (23)	119 (24)
2	79 (33)	76 (31)	155 (32)
3–5	100 (41)	113 (46)	213 (44)
Bulky disease (≥ 7 cm), n (%)	47 (19)	61 (25)	108 (22)

Data were previously published (Falchi et al. *Lancet*. 2026;407:161-173) and reproduced with permission under the terms of the CC BY 4.0 license.

Table 2: Treatment History Was Generally Balanced Across Epcoritamab+R^2 and R^2

	Epcoritamab+R^2 (N = 243)	R^2 (N = 245)	Overall (N = 488)
Median time from initial diagnosis to randomization, years (range)	4.5 (0.2, 30.3)	5.3 (0.1, 43.0)	5.0 (0.1, 43.0)
Number of prior lines of therapy, median (range)	1 (1, 7)	1 (1, 6)	1 (1, 7)
1, n (%)	145 (60)	141 (58)	286 (59)
2, n (%)	58 (24)	61 (25)	119 (24)
≥ 3, n (%)	40 (16)	43 (18)	83 (17)
Prior anti-CD20 antibody, n (%)	243 (100)	245 (100)	488 (100)
Prior anti-CD20 antibody containing chemotherapy, n (%)	239 (98)	240 (98)	479 (98)
Prior bendamustine in last line, n (%)	53 (22)	47 (19)	100 (20)
Prior R^2, n (%)	8 (3)	9 (4)	17 (3)
POD24,[a] n (%)	106 (44)	93 (38)	199 (41)
Refractory to 1L therapy, n (%)	86 (35)	81 (33)	167 (34)
Refractory to anti-CD20 antibody, n (%)	104 (43)	103 (42)	207 (42)
Refractory to last line of therapy, n (%)	84 (35)	82 (33)	166 (34)
Double refractory[b]	91 (37)	91 (37)	182 (37)

Data were previously published (Falchi et al. *Lancet*. 2026;407:161-173) and reproduced with permission under the terms of the CC BY 4.0 license.
[a]POD24 is defined as progression of disease ≤ 2 years from the date of initiation of frontline therapy. [b]Double refractory is refractory to prior anti-CD20 therapy and prior alkylator therapy.

Figure 1: Epcoritamab+R^2 Resulted in Higher Response Rates Versus R^2

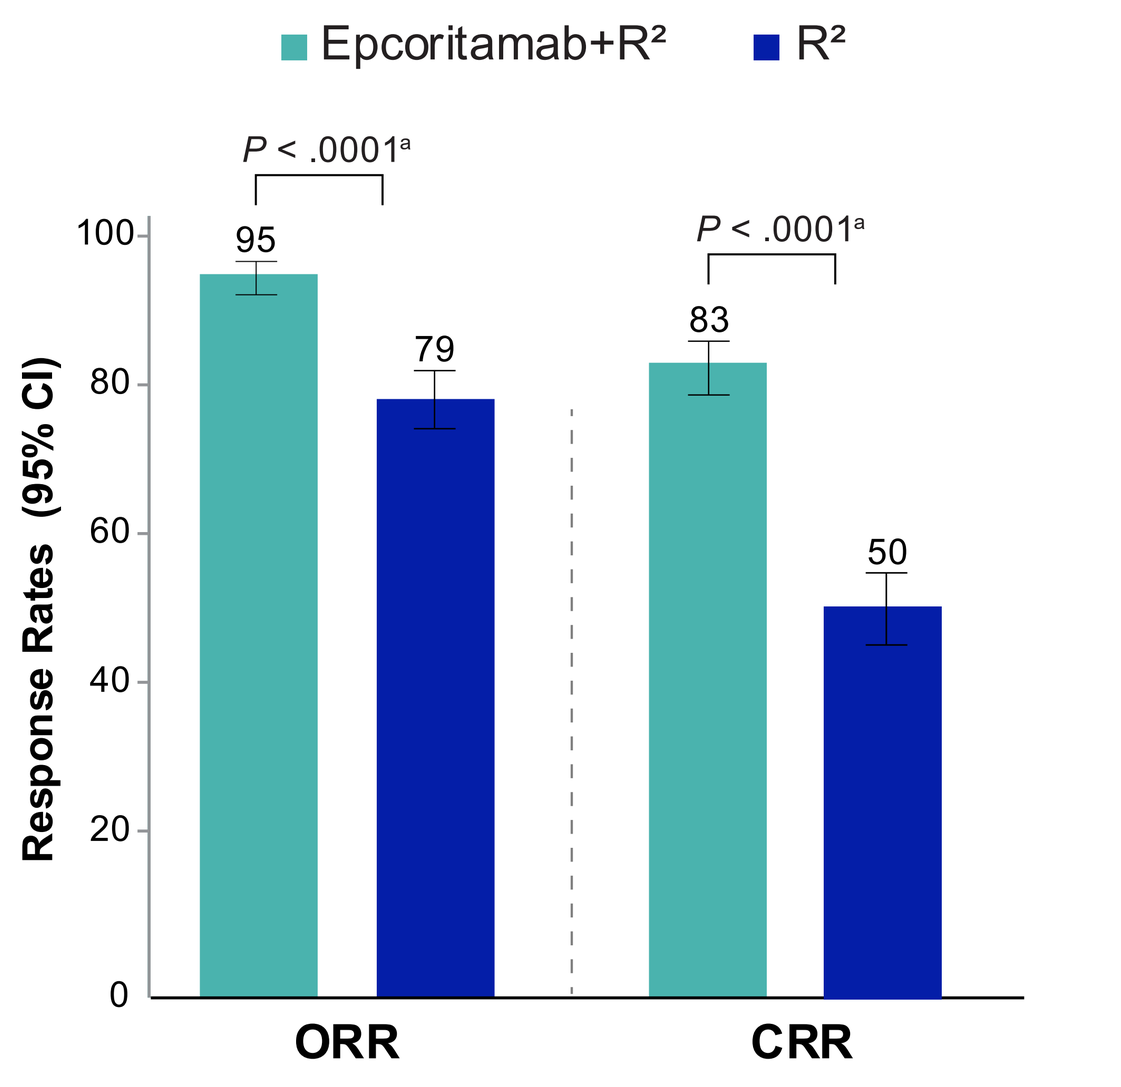
Epcoritamab+R²
R²
P < .0001ᵃ
P < .0001ᵃ
95
79
83
50
Response Rates (95% CI)
100
80
60
40
20
0
ORR
CRR

	Epcoritamab+R^2 (N = 243)	R^2 (N = 245)
ORR, n (%)	231 (95)	194 (79)
CRR, n (%)	201 (83)	122 (50)
PR, n (%)	30 (12)	72 (29)
SD, n (%)	1 (< 1)	17 (7)
PD, n (%)	7 (3)	16 (7)
NE,[b] n (%)	4 (2)	18 (7)

Data were previously published (Falchi et al. *Lancet*. 2026;407:161-173) and reproduced with permission under the terms of the CC BY 4.0 license.
The first planned interim analysis (January 10, 2025) achieved statistical significance for ORR (N = 232; 95.7% vs 81.0%; P < 0.0001, with a 1-sided significance level of 0.005) and CR (74.5% vs 43.3%; P < 0.0001, with a 1-sided significance level of 0.025).
[a]Nominal P value by stratified Cochran-Mantel-Haenszel method. [b]Patients with no post-baseline disease assessment were also included.

- DOR and DOCR were improved with epcoritamab+R^2 (Supplemental Materials accessible via the QR code)

Figure 2: Epcoritamab+R^2 Resulted in Superior PFS per IRC With 79% Risk Reduction

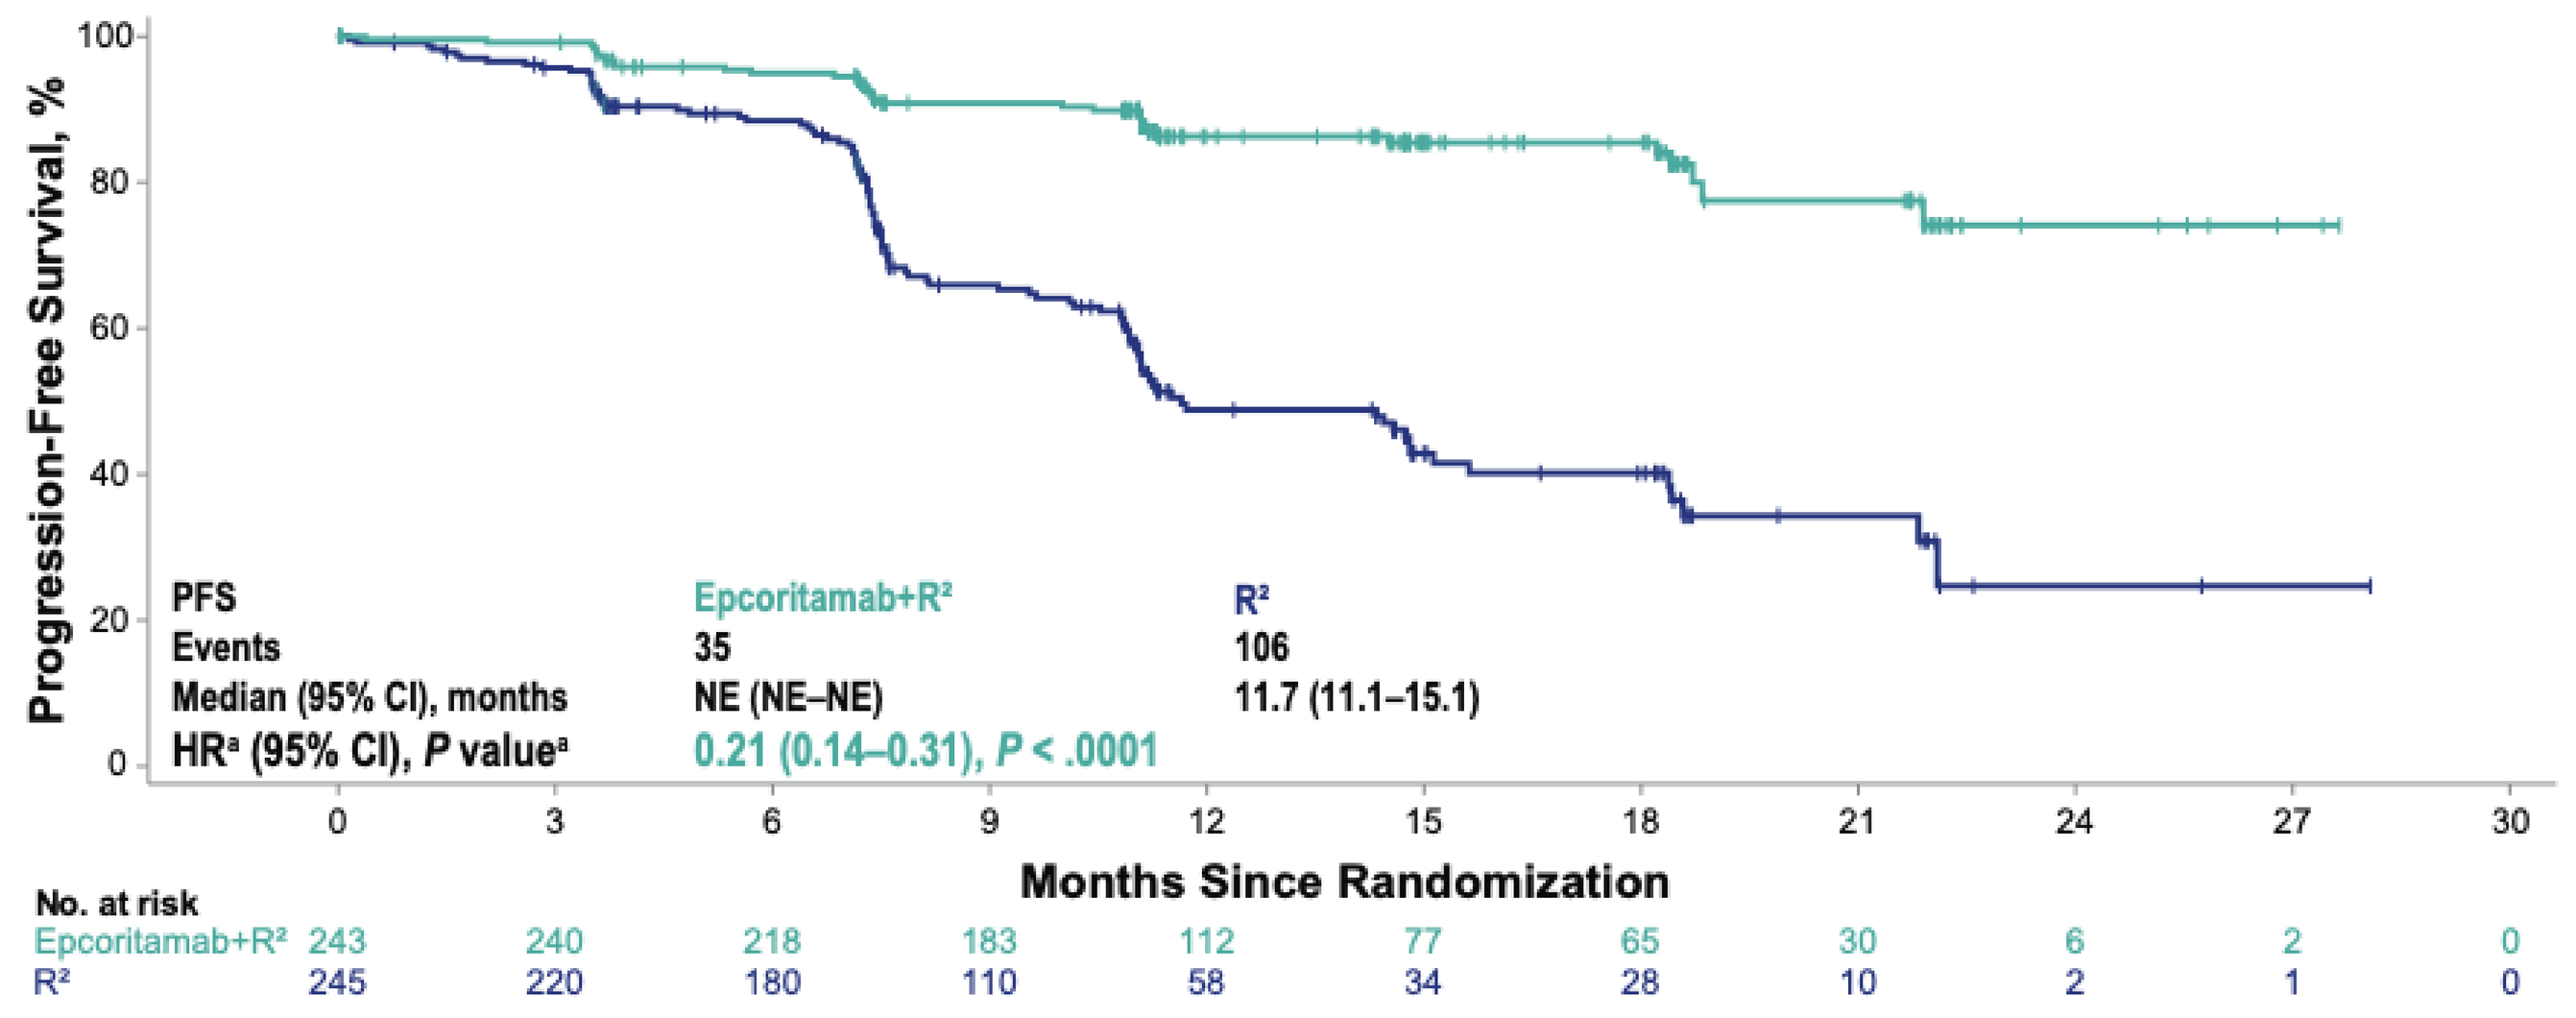
Progression-Free Survival, %
100
80
60
40
20
0
PFS | Epcoritamab+R² | R²
Events | 35 | 106
Median (95% CI), months | NE (NE–NE) | 11.7 (11.1–15.1)
HRᵃ (95% CI), P valueᵃ | 0.21 (0.14–0.31), P < .0001
0 3 6 9 12 15 18 21 24 27 30
Months Since Randomization
No. at risk
Epcoritamab+R² 243 240 218 183 112 77 65 30 6 2 0
R² 245 220 180 110 58 34 28 10 2 1 0

Data were previously published (Falchi et al. *Lancet*. 2026;407:161-173) and reproduced with permission under the terms of the CC BY 4.0 license.
Median follow-up for PFS: epcoritamab+R^2 (14.4m), R^2 (11.5m). The first planned interim analysis (January 10, 2025) achieved statistical significance on PFS, HR 0.21 (95% CI 0.13, 0.33) P < 0.0001, with a 1-sided significance level of 0.0023.
[a]Nominal P value is based on stratified log-rank test. Hazard ratio is estimated using stratified Cox proportional hazards model. This analysis was performed on the 78% information fraction.

- Concordance rate was 94% for PFS between IRC and investigator assessment
- The estimated 16-month PFS was 85.5% (95% CI: 79.7, 89.7) for epcoritamab+R^2 and 40.2% (95% CI: 31.8, 48.4) for R^2
- PFS showed trends in favor of epcoritamab+R^2 for all prespecified subgroups (Supplemental Materials)
- OS showed a positive trend with epcoritamab+R^2 (Supplemental Materials)

Figure 3: Epcoritamab+R^2 Extended Time to Next Treatment

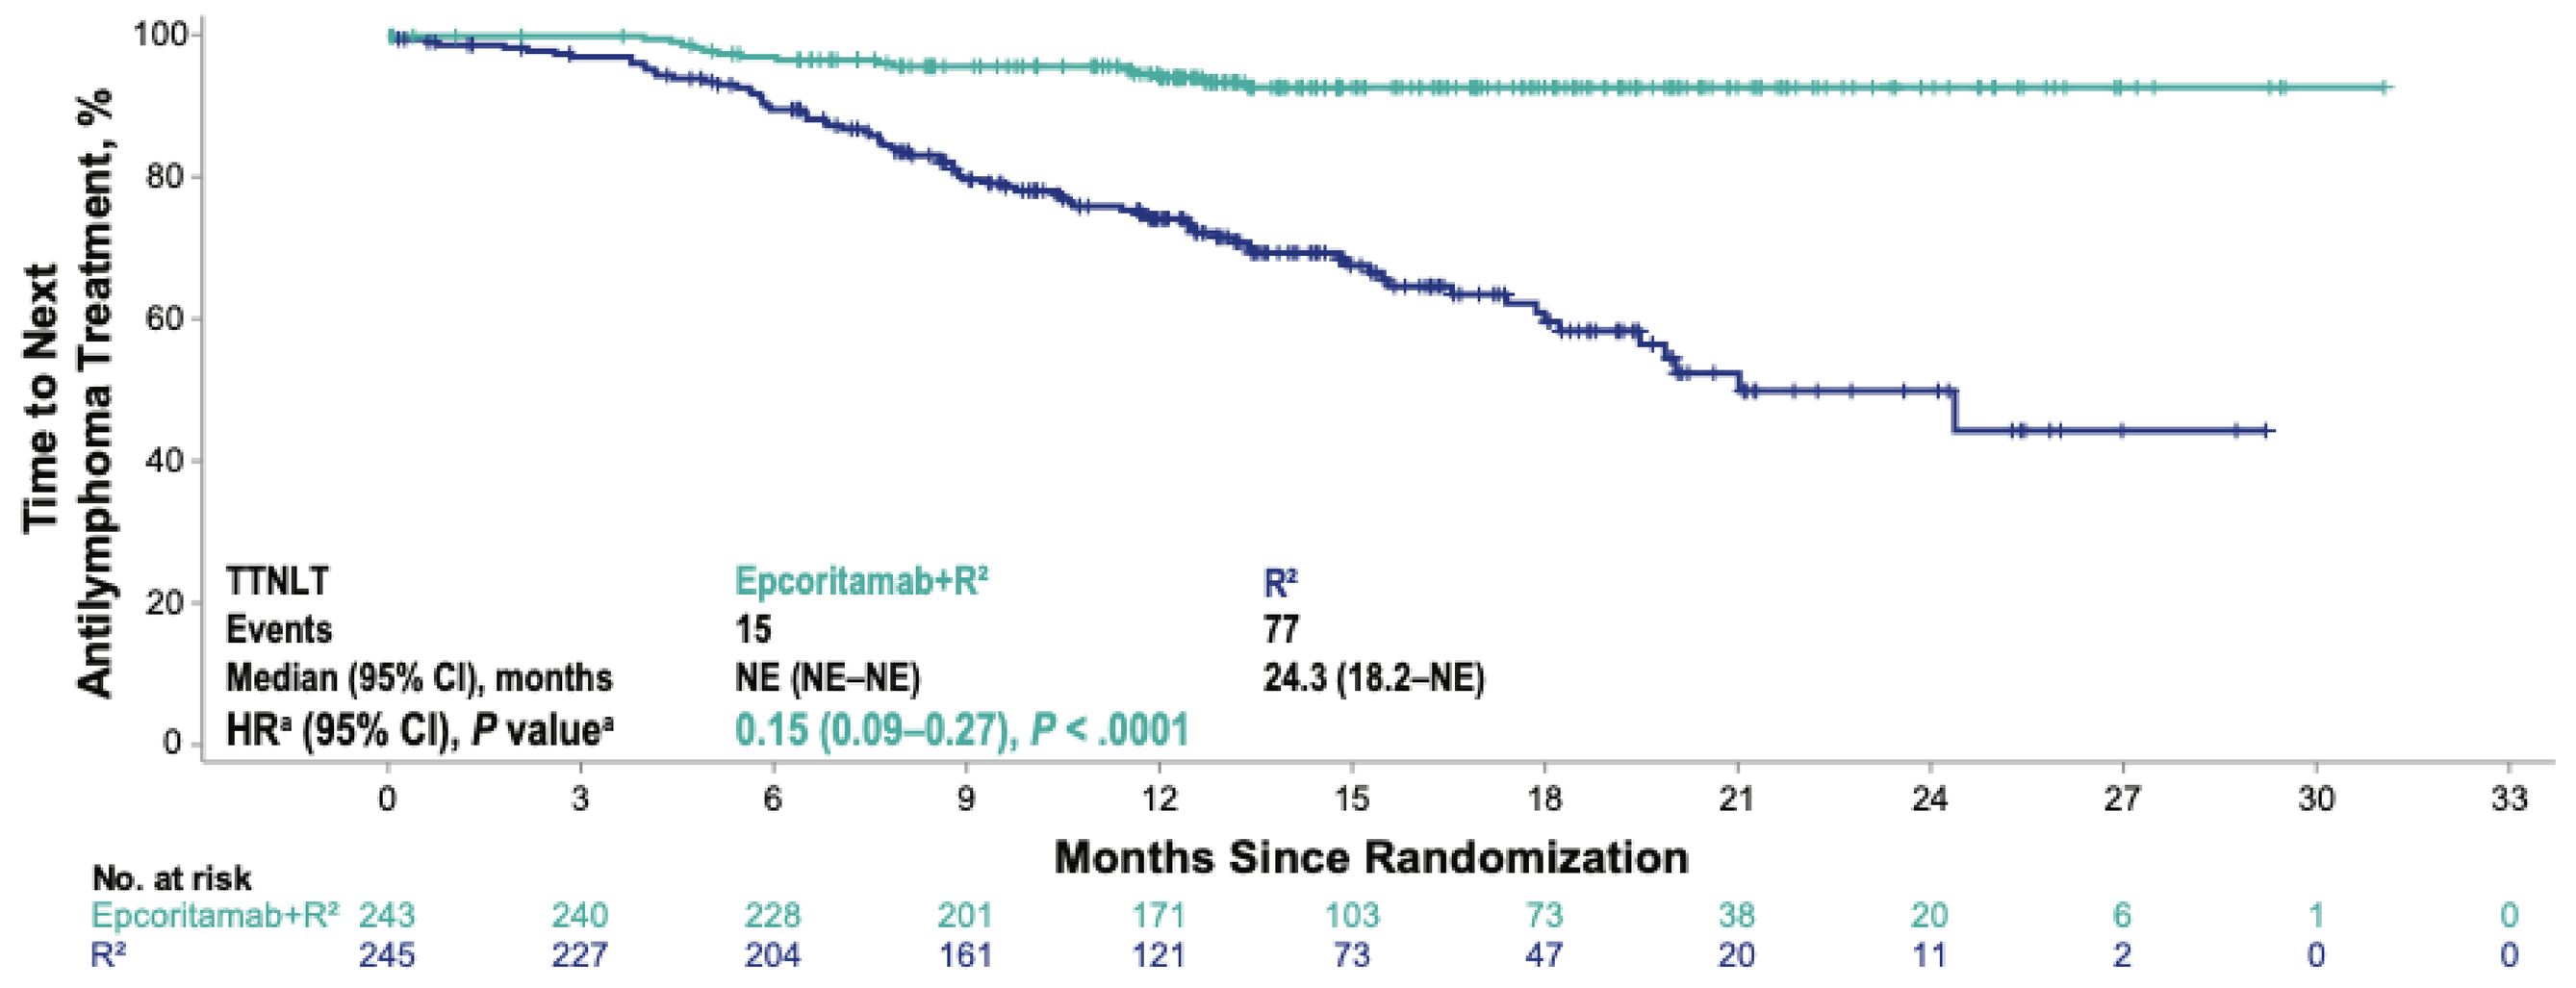
Time to Next Antilymphoma Treatment, %
100
80
60
40
20
0
TTNLT | Epcoritamab+R² | R²
Events | 15 | 77
Median (95% CI), months | NE (NE–NE) | 24.3 (18.2–NE)
HRᵃ (95% CI), P valueᵃ | 0.15 (0.09–0.27), P < .0001
0 3 6 9 12 15 18 21 24 27 30 33
Months Since Randomization
No. at risk
Epcoritamab+R² 243 240 228 201 171 103 73 38 20 6 1 0
R² 245 227 204 161 121 73 47 20 11 2 0 0

Data were previously published (Falchi et al. *Lancet*. 2026;407:161-173) and reproduced with permission under the terms of the CC BY 4.0 license.
Median follow-up for TTNLT: epcoritamab+R^2 (14.6m), R^2 (14.1m). TTNLT results are for descriptive purposes only.
[a]Nominal P value is based on stratified log-rank test. Hazard ratio is estimated using stratified Cox proportional hazards model.

- At 16 months, 92.8% of patients treated with epcoritamab+R^2 remained free from new antilymphoma treatment compared with 64.9% of patients treated with R^2

Table 3: Manageable AEs With No New Safety Signals

	Epcoritamab+R^2 (N = 243)		R^2 (N = 238)	
Adverse Event, n (%)	Any Grade	Grade ≥ 3	Any Grade	Grade ≥ 3
Any adverse event	242 (100)	219 (90)	235 (99)	161 (68)
Serious adverse event	135 (56)	–	69 (29)	–
Adverse event leading to treatment discontinuation	46 (19)	–	29 (12)	–
Epcoritamab	21 (9)	–	–	–
Rituximab	7 (3)	–	12 (5)	–
Lenalidomide	45 (19)	–	29 (12)	–
Adverse event of clinical interest > 20%[a,b]				
Infections[c]	188 (77)	81 (33)	125 (53)	37 (16)
Neutropenia	180 (74)	167 (69)	123 (52)	100 (42)
Cytokine release syndrome	85 (35)	–	1 (< 1)	–
Anemia	68 (28)	19 (8)	41 (17)	11 (5)
Thrombocytopenia	67 (28)	23 (9)	44 (18)	15 (6)
Pyrexia	58 (24)	1 (< 1)	33 (14)	3 (1)
Rash	58 (24)	19 (8)	53 (22)	9 (4)
COVID-19	54 (22)	7 (3)	32 (13)	4 (2)

Data were previously published (Falchi et al. *Lancet*. 2026;407:161-173) and reproduced with permission under the terms of the CC BY 4.0 license.
[a]Neutropenia, anemia, pyrexia, rash and COVID-19 are grouped terms comprising multiple clinically related Preferred Terms. [b]This includes the AESI of CRS.
[c]Events were in the MedDRA system organ class "Infections and Infestations." No grade 5 infections were reported.

- Neutropenia was manageable and few patients discontinued any study drug (epcoritamab+R^2, 3%; R^2, 2%)
 - Incidence of febrile neutropenia: epcoritamab+R^2, 6%; R^2, 3%
- Fatal adverse events were rare (epcoritamab+R^2, 2%; R^2, 4%)
- Despite higher rates of AEs in the epcoritamab+R^2 arm, most patients completed the prescribed regimen (median relative dose intensity ≥ 90% for epcoritamab+R^2)

Figure 4: Infection Risk Peaked in Cycles 1 and 2, Decreased Thereafter, and Stabilized After Cycle 4

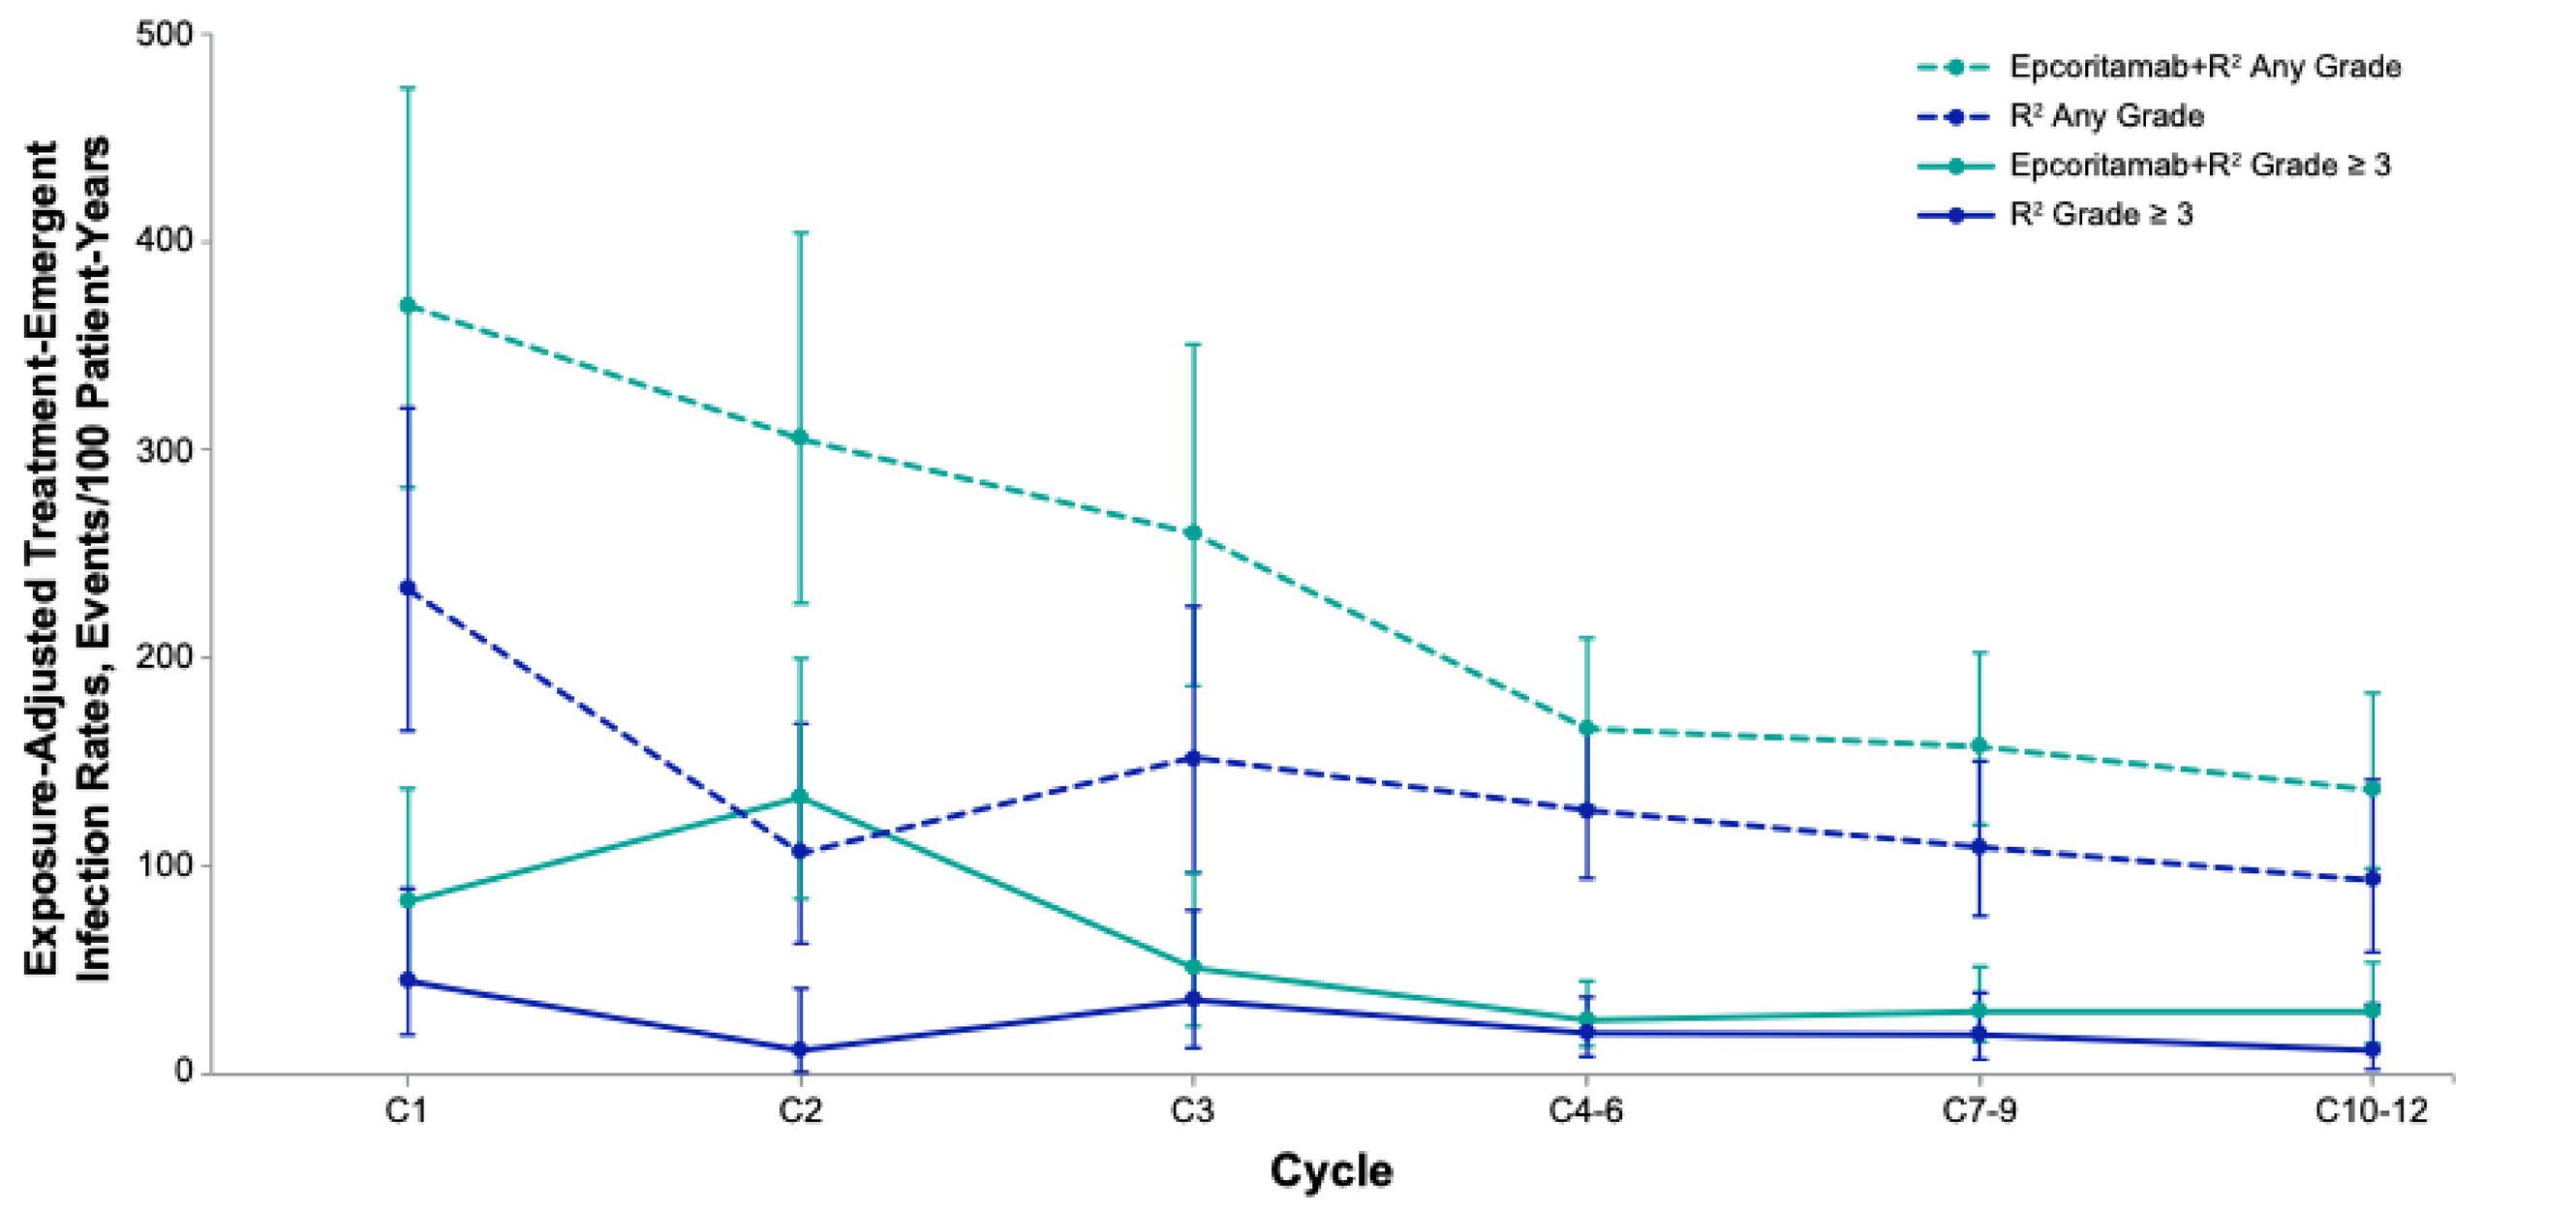
Epcoritamab+R² Any Grade
R² Any Grade
Epcoritamab+R² Grade ≥ 3
R² Grade ≥ 3
Exposure-Adjusted Treatment-Emergent Infection Rates, Events/100 Patient-Years
500
400
300
200
100
0
C1
C2
C3
C4-6
C7-9
C10-12
Cycle

EAIR (per 100 PY) = patients with ≥ 1 infection event ÷ patient-years at risk × 100, calculated separately by infection category and cycle group. Risk time was censored at the earliest of first qualifying infection event, death, end of the TEAE observation window, or last known alive date/data cutoff.

- Grade ≥ 3 exposure-adjusted treatment-emergent infection rates with epcoritamab+R^2 declined after cycle 2 and remained stable from cycle 4 onward, approaching rates observed with R^2 alone
- Infections were manageable and few patients discontinued any study drug (epcoritamab+R^2, 6%; R^2, 1%)

Table 4: CRS Was Low Grade With Epcoritamab+R^2

	Epcoritamab+R^2 2-SUD N = 110	Epcoritamab+R^2 3-SUD[a] N = 133
CRS, n (%)	50 (45)	35 (26)
CRS grade, n (%)		
1	40 (36)	28 (21)
2	10 (9)	7 (5)
CRS signs and symptoms, n (%)*		
Fever	49 (98)	33 (94)
Hypotension	9 (18)	6 (17)
Hypoxia	1 (2)	2 (6)
Time to first CRS onset from first full dose, days, median (range)	1 (< 1, 6)	1.5 (< 1, 10)
Time to CRS resolution, days, median (range)	1 (< 1, 12)	1 (< 1, 26)
CRS interventions, n (%)[a]		
Treated with tocilizumab	12 (24)	9 (26)
Treated with corticosteroid	23 (46)	13 (37)

Data were previously published (Falchi et al. *Lancet*. 2026;407:161-173) and reproduced with permission under the terms of the CC BY 4.0 license.
[a]Of patients who had CRS.

- Hydration and dexamethasone were utilized for CRS prophylaxis
- One event of ICANS was observed and was grade 1
- No discontinuations due to CRS and ICANS. All events resolved
- No events of CTLS were reported

Figure 5: CRS Timing Was Predictable With Epcoritamab+R^2

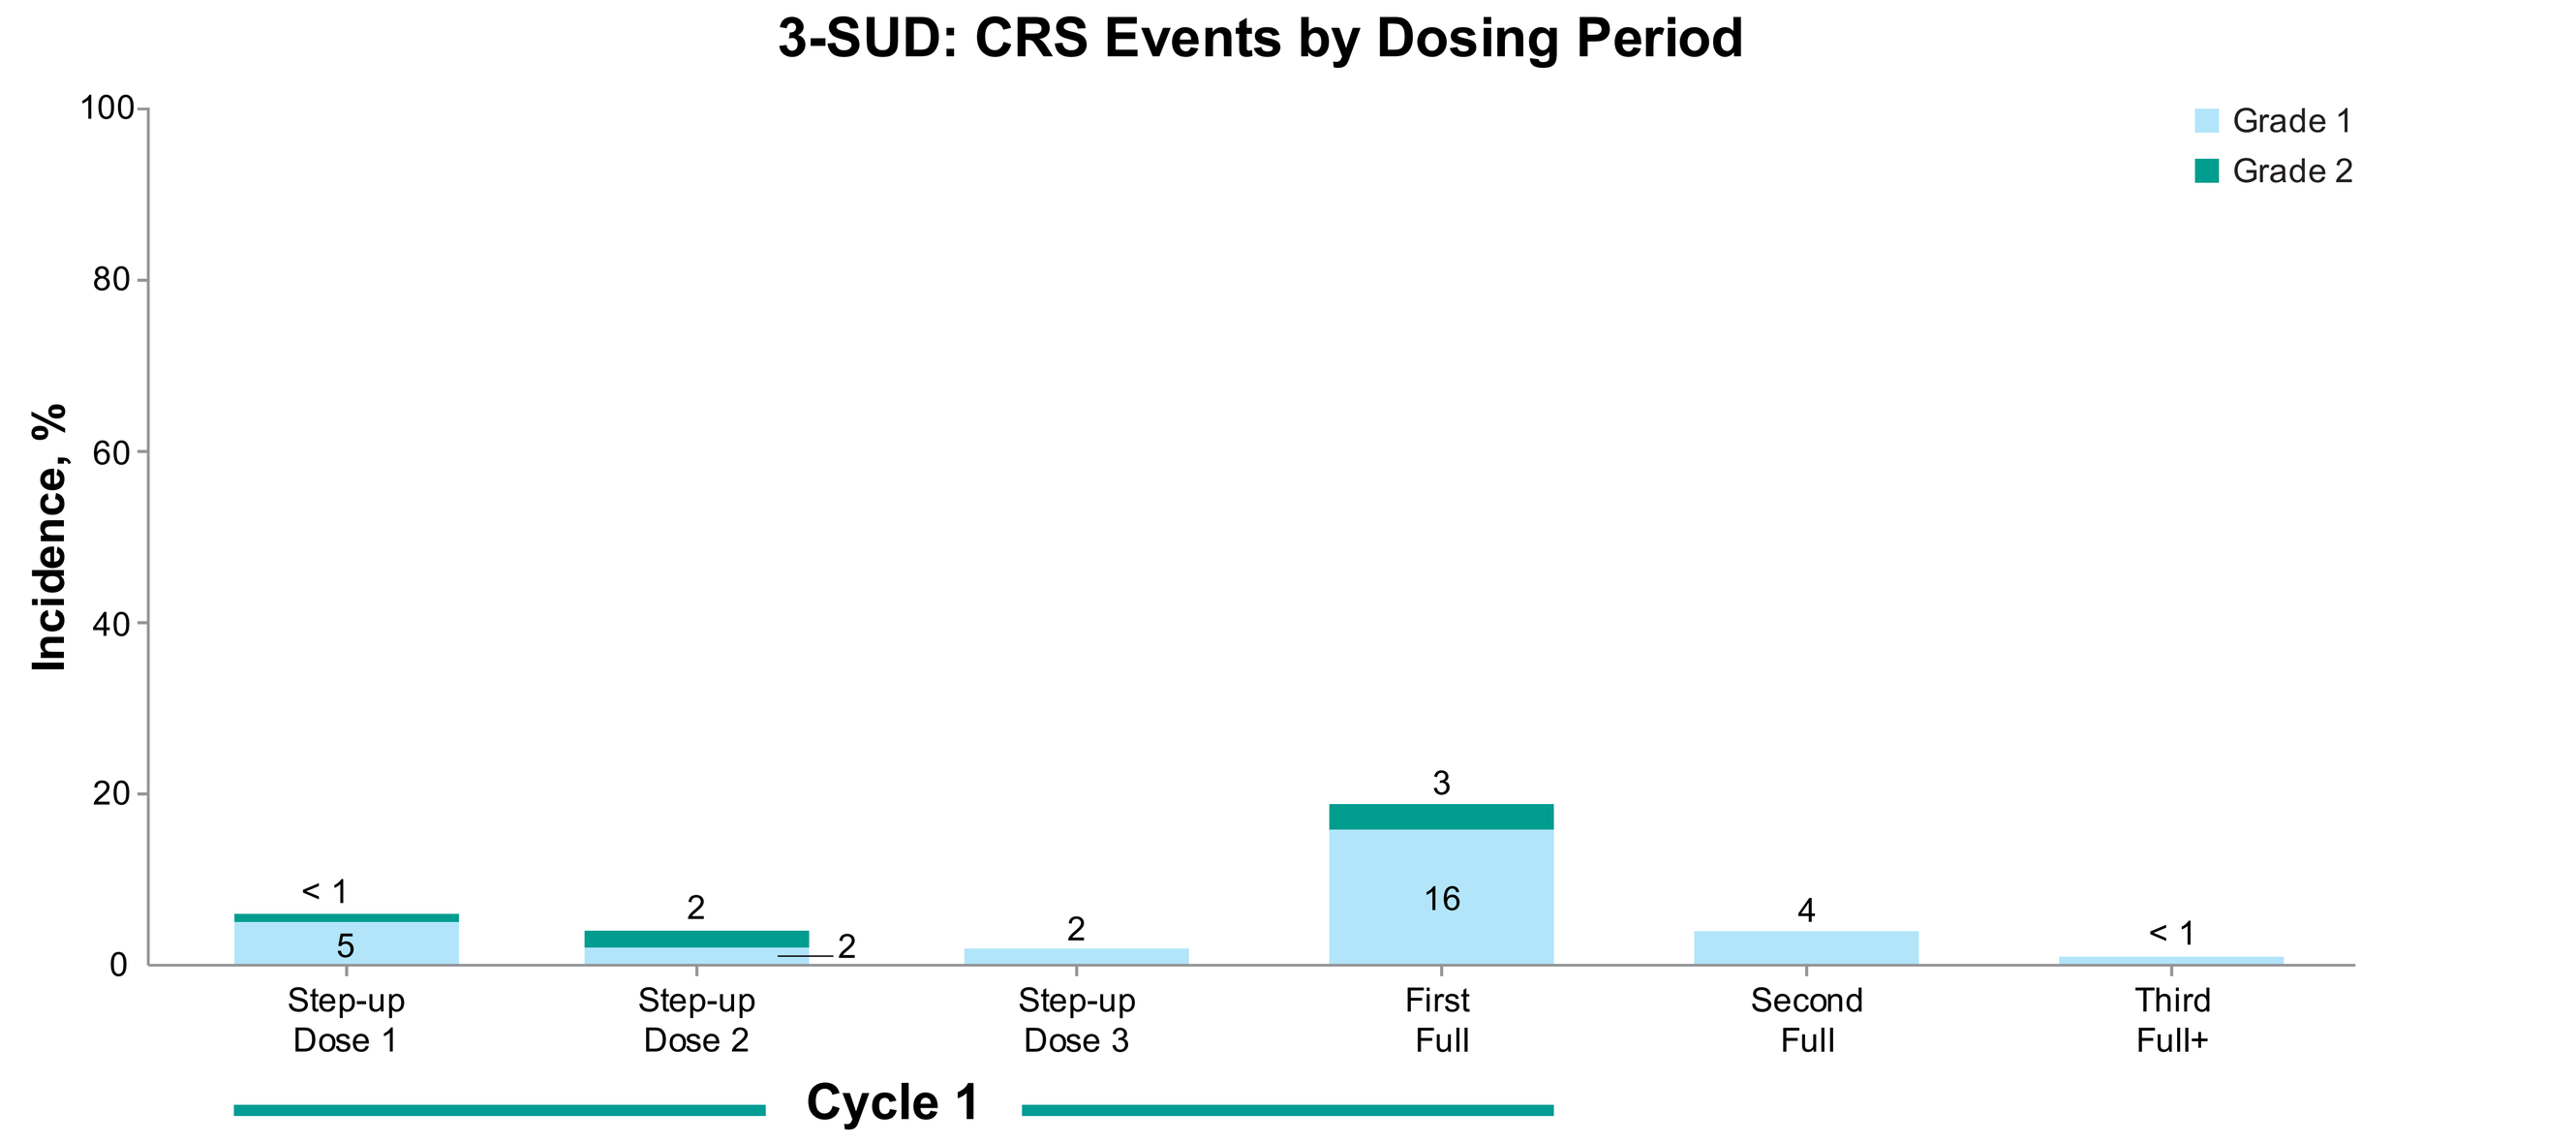
3-SUD: CRS Events by Dosing Period
Grade 1
Grade 2
Incidence, %
100
80
60
40
20
0
< 1
5
2
2
2
3
16
4
< 1
Step-up Dose 1
Step-up Dose 2
Step-up Dose 3
First Full
Second Full
Third Full+
Cycle 1

Data were previously published (Falchi et al. *Lancet*. 2026;407:161-173) and reproduced with permission under the terms of the CC BY 4.0 license.
[a]The 3-SUD regimen was implemented based off the EPCORE NHL-1 FL trial (NCT03625037).[11]