A Phase I Study of TP-3654, an Oral PIM Kinase Inhibitor, in Patients With Intermediate-2 or High-Risk Primary or Secondary Myelofibrosis (MF)

MPN-161

Claudia Lebedinsky[1], Sujan Kabir[1], Steven Warner[2], Golam Mohi[3], Huyuan Yang[1], Jian Mei[1], Edgar Braendle[1]

[1]Sumitomo Dainippon Pharma Oncology, Inc., Cambridge, MA; [2]Sumitomo Dainippon Pharma Oncology, Inc., Lehi, UT; [3]Department of Biochemistry and Molecular Genetics, University of Virginia School of Medicine, Charlottesville, VA

Background

- Myelofibrosis (MF) is characterized by bone marrow fibrosis (BMF), ineffective hematopoiesis, splenomegaly, and debilitating symptoms
- The current standard of care treatment options, Janus kinase (JAK) inhibitors ruxolitinib and fedratinib, are indicated for patients with MF who have platelet counts ≥50 × 10^9/L
 - JAK inhibitors reduced spleen volume and improved constitutional symptoms,[1–4] but have limited disease-modifying activity in reducing the mutant allele burden or BMF[1,5]
 - In addition, disease-related thrombocytopenia or JAK inhibitors-related thrombocytopenia will lead to dose reduction, which further limits potential benefit[6]
 - Patients with MF do not have any approved treatment options after progression following JAK inhibitor
- TP-3654 is an orally delivered Proviral Integration site for Moloney murine leukemia virus (PIM) kinase inhibitor, with highest potency against PIM-1
 - PIM kinases are serine/threonine kinases involved in several signaling pathways important in myeloproliferative neoplasms (MPN) like MF
 - PIM kinases are downstream targets of the JAK/signal transducers and activators of transcription (STAT) signaling pathway leading to cell survival and drug resistance[7,8] (**Figure 1**)

Figure 1. Role of PIM Kinase in Cancer[a]

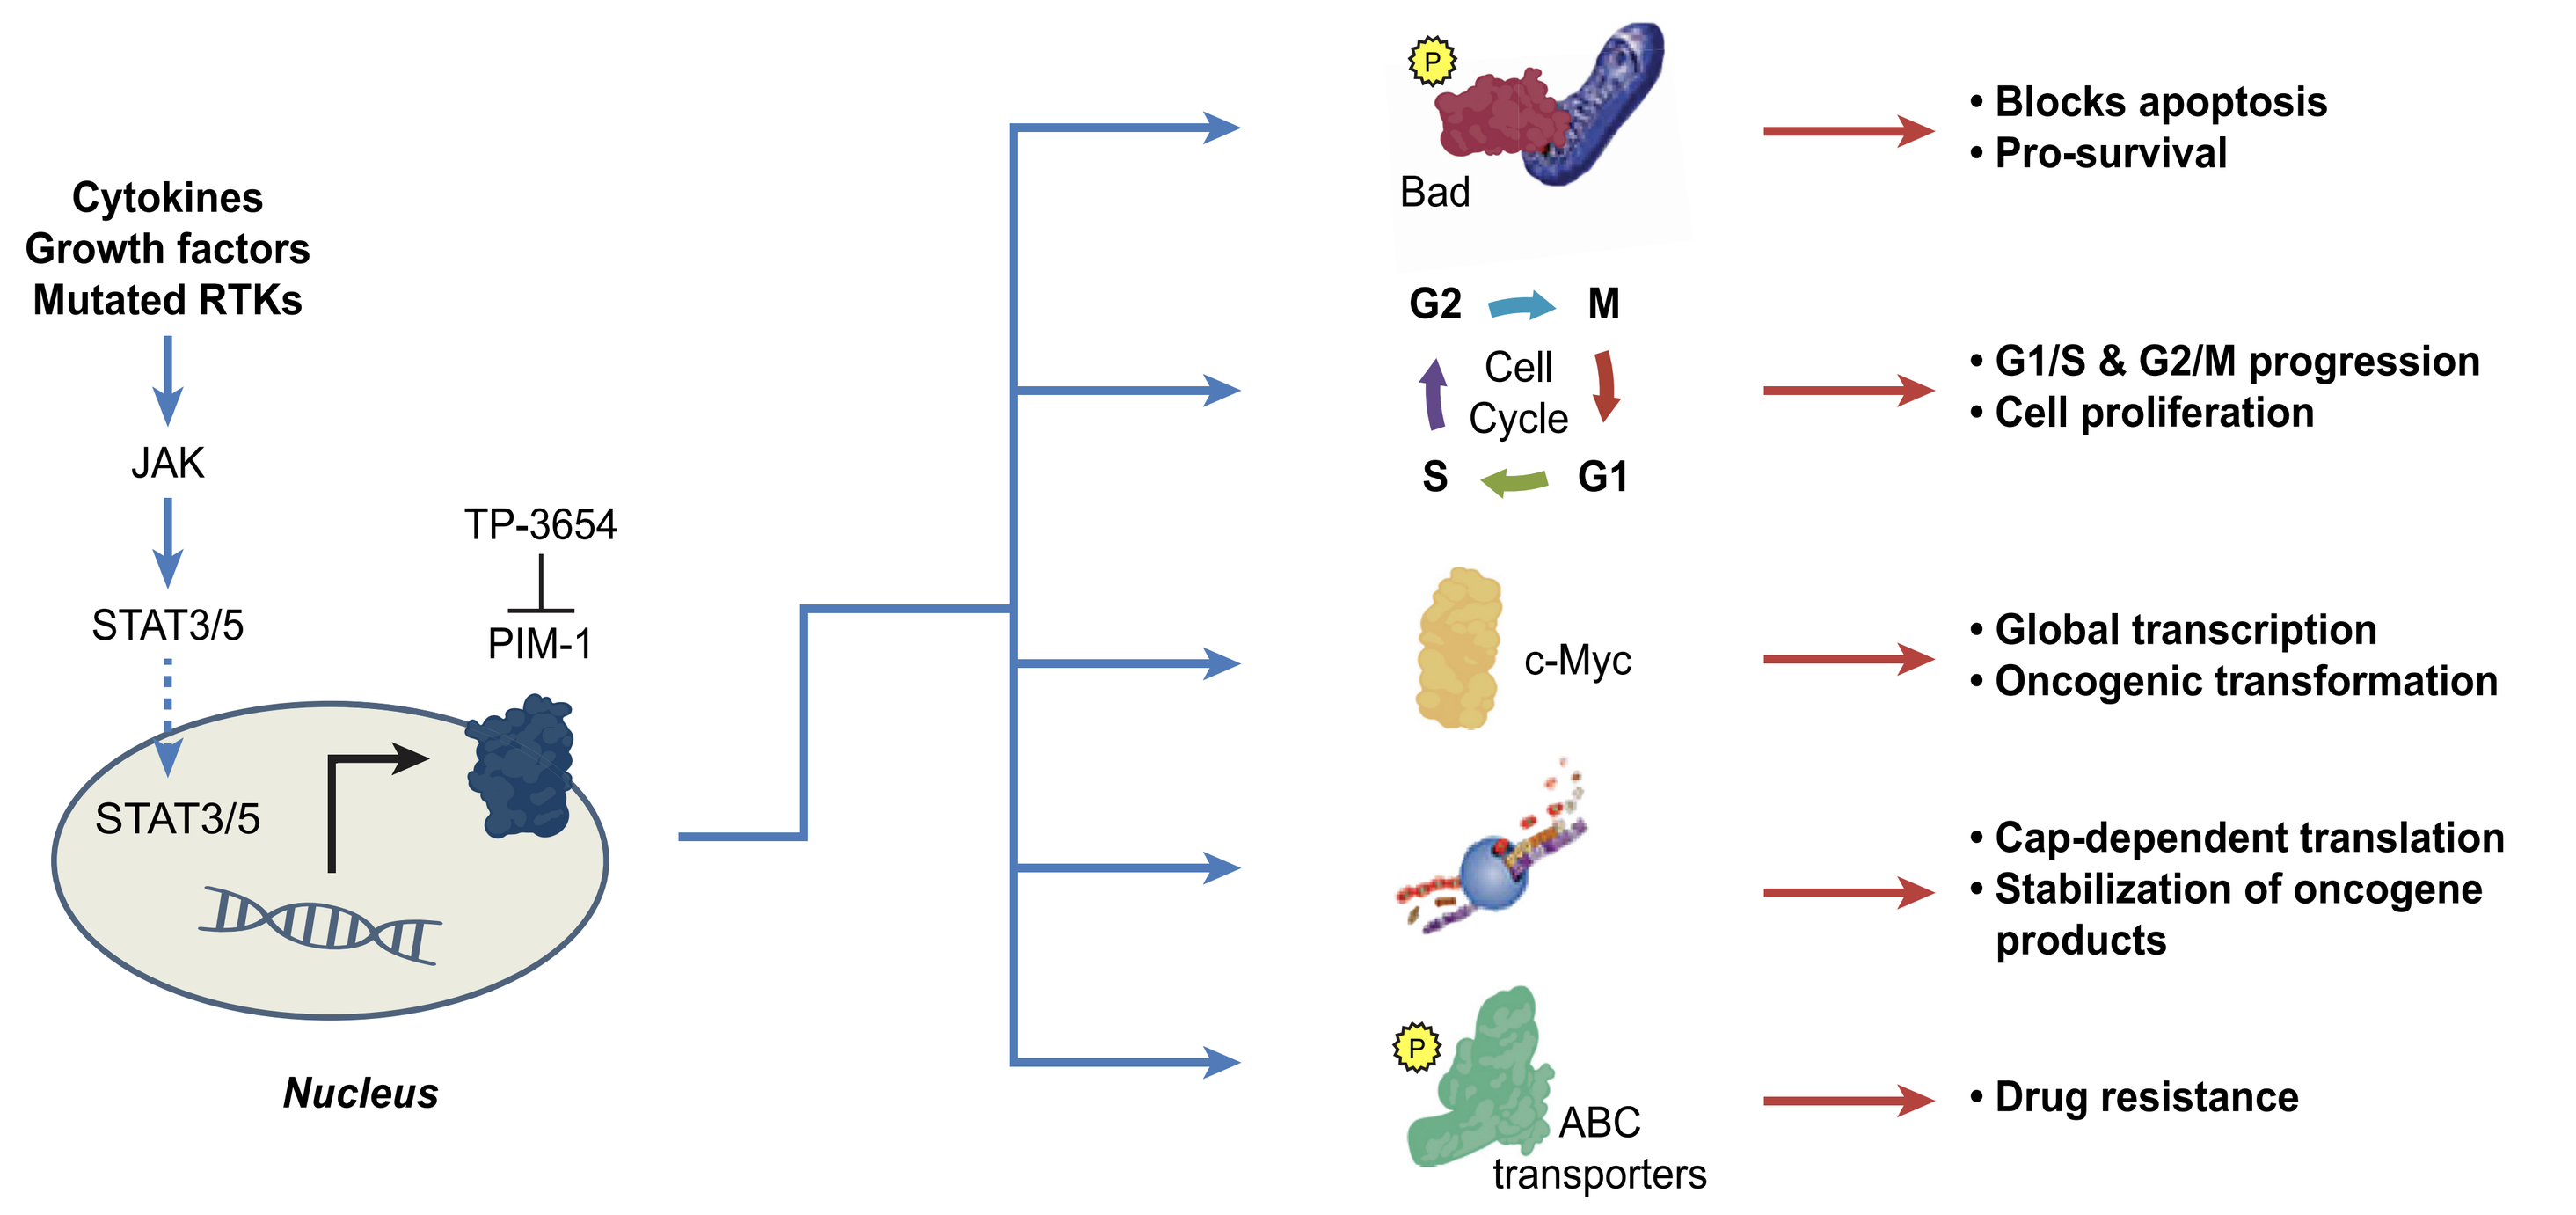
Cytokines
Growth factors
Mutated RTKs
JAK
STAT3/5
TP-3654
PIM-1
STAT3/5
Nucleus
Bad
• Blocks apoptosis
• Pro-survival
G2
M
Cell Cycle
S
G1
• G1/S & G2/M progression
• Cell proliferation
c-Myc
• Global transcription
• Oncogenic transformation
• Cap-dependent translation
• Stabilization of oncogene products
ABC transporters
• Drug resistance

[a]Adapted with permission from Garrido-Laguna I, et al. ASCO 2020. Poster 3586.

ABC, ATP-binding cassette; Bad, BCL-2 associated agonist of cell death; JAK, Janus kinase; Myc, myelocytomatosis; PIM, proviral integration site of moloney murine leukemia virus; RTK, receptor tyrosine kinase; STAT, signal transducer and activator of transcription.

- PIM-1 expression is increased in granulocytes and peripheral blood mononuclear cells in patients with MPN (polycythemia vera, essential thrombocythemia and MF) compared with healthy controls, irrespective of JAK2, MPL or CALR mutations[9–11]
 - PIM kinases promote cell cycle progression by several mechanisms, including PIM-1 that phosphorylates CDC25A, leading to an increase in its phosphatase activity[12]
 - Overexpression of PIM kinases has also been linked to ruxolitinib drug resistance in MF[9]
- PIM-1 kinase inhibitors may be a treatment for MPN
- TP-3654, alone and in combination with ruxolitinib, reduces spleen size and BMF in JAK2V617F and MPLW515L murine MF models (**Figure 2**)[10]

Figure 2. TP-3654 Reduce Spleen Size and Bone Marrow Fibrosis[a]

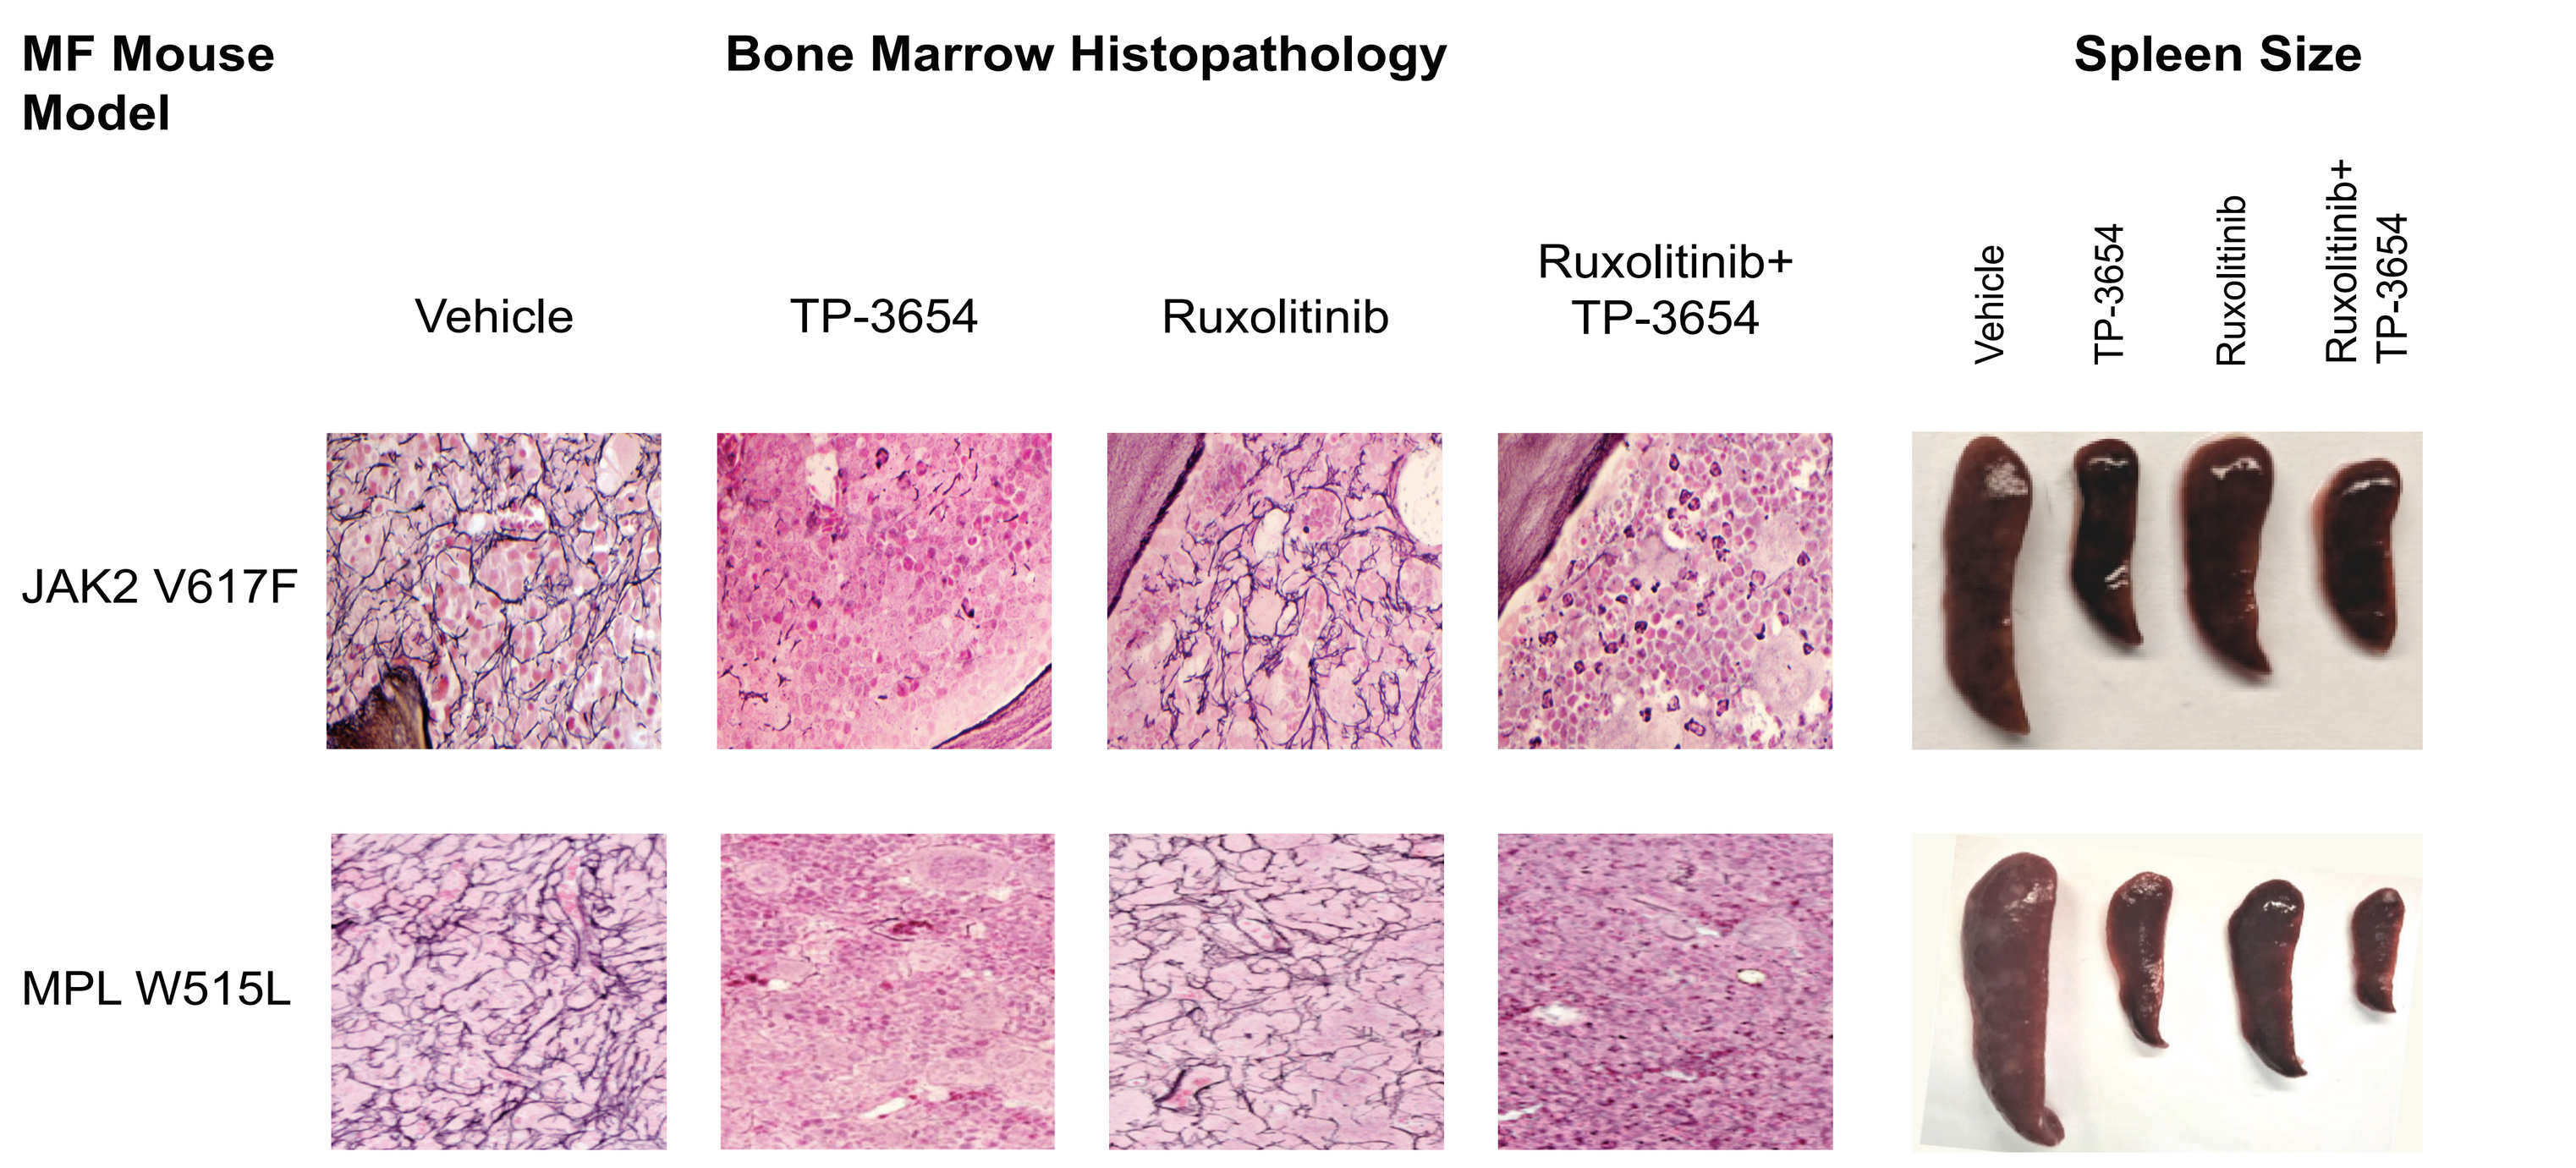
MF Mouse Model
Bone Marrow Histopathology
Spleen Size
Vehicle
TP-3654
Ruxolitinib
Ruxolitinib+ TP-3654
Vehicle
TP-3654
Ruxolitinib
Ruxolitinib+ TP-3654
JAK2 V617F
MPL W515L

JAK, Janus kinase; MF, myelofibrosis; MPL, myeloproliferative leukemia.

[a]Bone marrow histology (reticulin staining) and spleen images from Dr. Mohi's Laboratory at the University of Virginia. Bone marrow histology images adapted with permission from Mohi G, et al. ASH 2018. Oral presentation, Abstract #54. https://doi.org/10.1182/blood-2018-99-119421.

Purpose

- To describe the safety, pharmacokinetics, and preliminary activity of TP-3654 in patients with intermediate-2 or high-risk MF who have been previously treated with a JAK inhibitor and have become intolerant, resistant, refractory, or are ineligible to be treated with a JAK inhibitor (NCT04176198)

Study Design

Study Treatment

- TP-3654 is administered orally daily in a 28-day dosing cycle (**Figure 3**)

Figure 3. Study Design

Phase 1 (Open-Label)

DIPSS Intermediate-2 or high-risk primary or secondary MF patients who have been previously treated with a JAK inhibitor and have become intolerant, resistant, refractory, or are ineligible to be treated with a JAK inhibitor

TP-3654 is taken orally each day

Approximate enrollment: 21 to 50 patients

Dose Escalation by BLRM

Each cohort will enroll 1–6 patients for safety evaluation

If a DLT is observed, ≥3 patients must be treated per cohort

Escalation to higher dose levels based on review of DLTs during the first cycle (28 days)

BLRM, Bayesian Logistic Regression Model; DIPSS, Dynamic International Prognostic Scoring System; DLT, dose limiting toxicity; JAK, Janus kinase; MF, myelofibrosis.

Table 1. Study Objectives and Endpoints

	Objectives	Endpoints
Primary	• Determine the MTD and/or identify the RP2D of TP-3654 • Determine the overall safety of TP-3654	• DLTs at escalated doses of TP-3654 • AEs as characterized by type, frequency, severity,[a] seriousness, and relationship to TP-3654
Secondary	• Establish the PK profile of orally administered TP-3654 • Assess patients for any evidence of preliminary activity of TP-3654 • Determine the cardiac safety of TP-3654	• PK parameters of TP-3654, including C_{max}, t_{max}, AUC, $t_{½}$, and others • Proportion of patients with ≥35% spleen volume reduction (SVR35) at 24 weeks post-treatment • Proportion of patients with ≥50% improvement in total symptom score (TSS50) at 24 weeks post-treatment by MFSAF v4.0 • Change in BMF in repeat biopsies[b] • Preliminary activity of TP-3654 per the revised IWG-MRT[13] response criteria • QT interval changes and morphology as assessed by Holter ECG monitoring
Exploratory	• Study potential pharmacodynamic markers of TP-3654	• Exploratory biomarkers in peripheral blood samples and bone marrow biopsy samples

[a]Graded in accordance with NCI CTCAE v5.0. [b]Per the European Consensus Fibrosis Guide.

AE, adverse event; AUC, area under the curve; BMF, bone marrow fibrosis; C_{max}, maximum concentration; CTCAE, Common Terminology Criteria for Adverse Events; DLT, dose-limiting toxicity; ECG, electrocardiogram; IWG-MRT, International Working Group for Myelofibrosis Research and Treatment; MFSAF, Myelofibrosis Symptom Assessment Form; MTD, maximum tolerated dose; NCI, National Cancer Institute; PK, pharmacokinetic; RP2D, recommended phase II dose; $t_{1/2}$, half-life; T_{max}, time to maximum concentration.

Statistical Design and Analysis

- Dose escalation will be performed based on the Bayesian logistic regression model (BLRM) with escalation with overdose control principle to control the risk of exposing patients to toxic doses
- A dose level will be considered safe if there is ≤25% probability of reaching a toxic dose (DLT rate >33%)
- The dose recommended by the BLRM method will serve as a guide and will be considered together with the understanding of clinical assessment of the toxicity information, pharmacokinetics, and biomarker data to determine the MTD or RP2D

Current Status

- The study is currently recruiting patients
- For more details about the study or to contact the Sponsor, please see: https://clinicaltrials.gov/ct2/show/NCT04176198

Table 2. Key Eligibility Criteria

Key Inclusion Criteria	Key Exclusion Criteria
• Confirmed pathologic diagnosis of primary MF or post-PV-MF/post-ET-MF (WHO diagnostic criteria), and intermediate-2 or high-risk based on DIPSS • Previously treated with a JAK inhibitor and are intolerant, resistant, refractory or lost response to the JAK inhibitor, or are ineligible to be treated with ruxolitinib or fedratinib • Grade ≥2 BMF as confirmed by BM biopsy ≤12 weeks prior to screening • **Platelet count ≥50 x 10^9/L** • Absolute neutrophil count ≥1 x 10^9/L • ECOG PS ≤2 • Adequate renal and hepatic function • Splenomegaly during screening (ie, splenic length ≥5 cm below the costal margin by palpation or spleen volume ≥450 cm^3 by MRI or CT scan) • ≥2 measurable symptoms (score ≥1) per the MFSAF, v4.0	• Splenic irradiation ≤6 months prior to screening or prior splenectomy • AML, MDS, or peripheral blasts ≥10% • Prior autologous SCT or allogeneic SCT • Eligible for allogeneic BMT or SCT ≤3 months after enrollment (not a candidate or not willing to undergo transplantation or a suitable donor not available are considered as transplant ineligible) • Received previous systemic antineoplastic therapy (including unconjugated therapeutic antibodies, toxin immunoconjugates, ESA, and alpha-interferon) or any experimental therapy within 14 days or 5 half-lives, whichever is longer, prior to first dose • Used hydroxyurea or anagrelide ≤24 hours prior to first dose

AML, acute myeloid leukemia; BM, bone marrow; BMF, bone marrow fibrosis; BMT, bone marrow transplant; CT, computerized tomography; DIPSS, Dynamic International Prognostic Scoring System; ECG, electrocardiogram; ECOG PS, Eastern Cooperative Oncology Group performance status; ESA, erythropoiesis-stimulating agent; ET, essential thrombocythemia; JAK, Janus kinase; MDS, myelodysplastic syndrome; MF, myelofibrosis; MFSAF, Myelofibrosis Symptom Assessment Form; MRI, magnetic resonance imaging; PV, polycythemia vera; SCT, stem cell transplant; WHO, World Health Organization.

References

1. Verstovsek S, et al. *N Engl J Med*. 2012;366:799–807. **2.** Cervantes F, et al. *Blood*. 2013;122:4047–53. **3.** Harrison C, et al. *N Engl J Med*. 2012;366:787–98. **4.** Davis KL, et al. *Adv Hematol.* 2015;2015:848473. **5.** Verstovsek S, et al. *J Hematol Oncol.* 2017;10(1):156. **6.** Ruxolitinib prescribing information. Incyte Corporation, Wilmington, DE, USA. January 2020. Accessed July 30, 2021. https://www.jakafi.com/pdf/prescribing-information.pdf. **7.** Zhang X, et al. *J Cancer Prev.* 2018;23:109–16. **8.** Mondello P, et al. *J Hematol Oncol*. 2014;7:95. **9.** Dutta A, et al. *Cancer Res.* 2018;78(13 Suppl): Abstract nr 1874 (AACR 2018). **10.** Data on file, Sumitomo Dainippon Pharma Oncology, Inc. **11.** Rampal R, et al. *Blood*. 2014;123:e123–33. **12.** Rampal RK, et al. *Clin Cancer Res*. 2021;27:3456–68. **13.** Tefferi A, e tal. *Blood 2013*; 122:1395–8.

Acknowledgements

This study was sponsored by Sumitomo Dainippon Pharma Oncology, Inc. The authors wish to thank the investigators, site coordinators, and the patients for participation in this study. Medical writing assistance was provided by Ashfield MedComms, an Ashfield Health Company, and funded by Sumitomo Dainippon Pharma Oncology, Inc.

Disclosures

CL, SK, SW, HY, JM, and EB are employees of Sumitomo Dainippon Pharma Oncology, Inc. GM received research funding from Tolero Pharmaceuticals, Inc. (now Sumitomo Dainippon Pharma Oncology, Inc.)

Society of Hematologic Oncology (SOHO) Ninth Hybrid Annual Meeting | September 8–11, 2021