MDS-420

Guillermo Garcia-Manero, ggarciam@mdanderson.org

Sabatolimab Plus Hypomethylating Agents (HMAs) in Patients With High-/Very High-risk Myelodysplastic Syndrome (HR/vHR-MDS) and Newly Diagnosed Acute Myeloid Leukemia (ND-AML): Subgroup Analysis of a Phase 1 Study

Guillermo Garcia-Manero,[1] Andrew H. Wei,[2] Kimmo Porkka,[3] Steve Knapper,[4] Elie Traer,[5] Sebastian Scholl,[6] Norbert Vey,[7] Martin Wermke,[8] Jeroen Janssen,[9] Rupa Narayan,[10] Sun Loo,[11] Natalia Tovar,[12] Mika Kontro,[3] Oliver Ottmann,[4] Purushotham Naidu,[13] Elena Orlando,[14] Nidhi Patel,[14] Jessica Makofske,[14] Fei Ma,[13] Na Zhang,[14] Anisa Mohammed,[13] Mikael L. Rinne,[14] Uma Borate,[5] Andrew M. Brunner[10]

[1]MD Anderson Cancer Center, Houston, TX, USA; [2]The Alfred Hospital and Monash University, Melbourne, Australia; [3]Helsinki University Hospital Comprehensive Cancer Center, Helsinki, Finland; [4]Cardiff University, Cardiff, UK; [5]Oregon Health & Science University, Portland, OR, USA; [6]University Hospital Jena, Jena, Germany; [7]Institut Paoli-Calmettes, Marseille, France; [8]University Hospital Dresden, Dresden, Germany; [9]Amsterdam University Medical Centers, location VUmc, Amsterdam, The Netherlands; [10]Massachusetts General Hospital, Boston, MA, USA; [11]The Alfred Hospital, Melbourne, Victoria, Australia; [12]Hospital Clinic, Barcelona, Spain; [13]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [14]Novartis Institutes for BioMedical Research, Cambridge, MA, USA.

Scan to obtain:
- Poster

http://novartis.medicalcongressposters.com/Default.aspx?doc=180c7

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

CONCLUSIONS

- Sabatolimab + HMA is well tolerated in MDS/AML
 - Very few patients had clinically significant treatment-related possible imAEs
- Sabatolimab + HMA demonstrated durable clinical benefit in patients with vHR/HR-MDS and ND-AML
 - vHR/HR-MDS, ORR: 58%; Median DOR: 16.1 months (95% CI, 6.7-NE)
 - ND-AML, ORR: 42.5%; Median DOR: 12.6 months (95% CI, 5.2-NE)
- Durable responses seen in patients with adverse risk mutations (TP53/RUNXL1/ASXL1) or poor cytogenetic risk
- Sabatolimab + HMA downregulates pro-inflammatory IL-1β in leukemic blasts and progenitors and upregulates IL-1β in immune myeloid cells
- Further Phase 2/3 studies of sabatolimab + HMA are underway as part of the STIMULUS immuno-myeloid clinical trial program

This study is sponsored by Novartis Pharmaceuticals Corporation.
Presented at the Society of Hematologic Oncology Ninth Hybrid Annual Meeting; September 8-11, 2021.

INTRODUCTION

TIM-3

- Involved in regulating innate and adaptive immune responses[1,2] (**Figure 1**)
- Aberrantly expressed on leukemic stem cells (LSCs) and blasts, but not on normal hematopoetic stem cells (HSCs),[1-5] making it a promising target in myelodysplastic syndrome (MDS) and acute myeloid leukemia (AML)[2,4,6]
- T-cell immunoglobin domain and mucin domain-3 (TIM-3)/galectin-9 interaction forms an autocrine stimulatory loop promoting LSC self-renewal[2,7,8]

Sabatolimab

- Binds TIM-3 on immune cells, enhancing antileukemic immune activation and phagocytic killing of LSCs and blasts[9-12] (**Figure 2**)
- Directly targets TIM-3 on LSCs, inhibiting TIM-3/galectin-9–driven self-renewal[9,10]

Figure 1. TIM-3 is an Inhibitory Receptor Expressed on Immune and Leukemic Cells

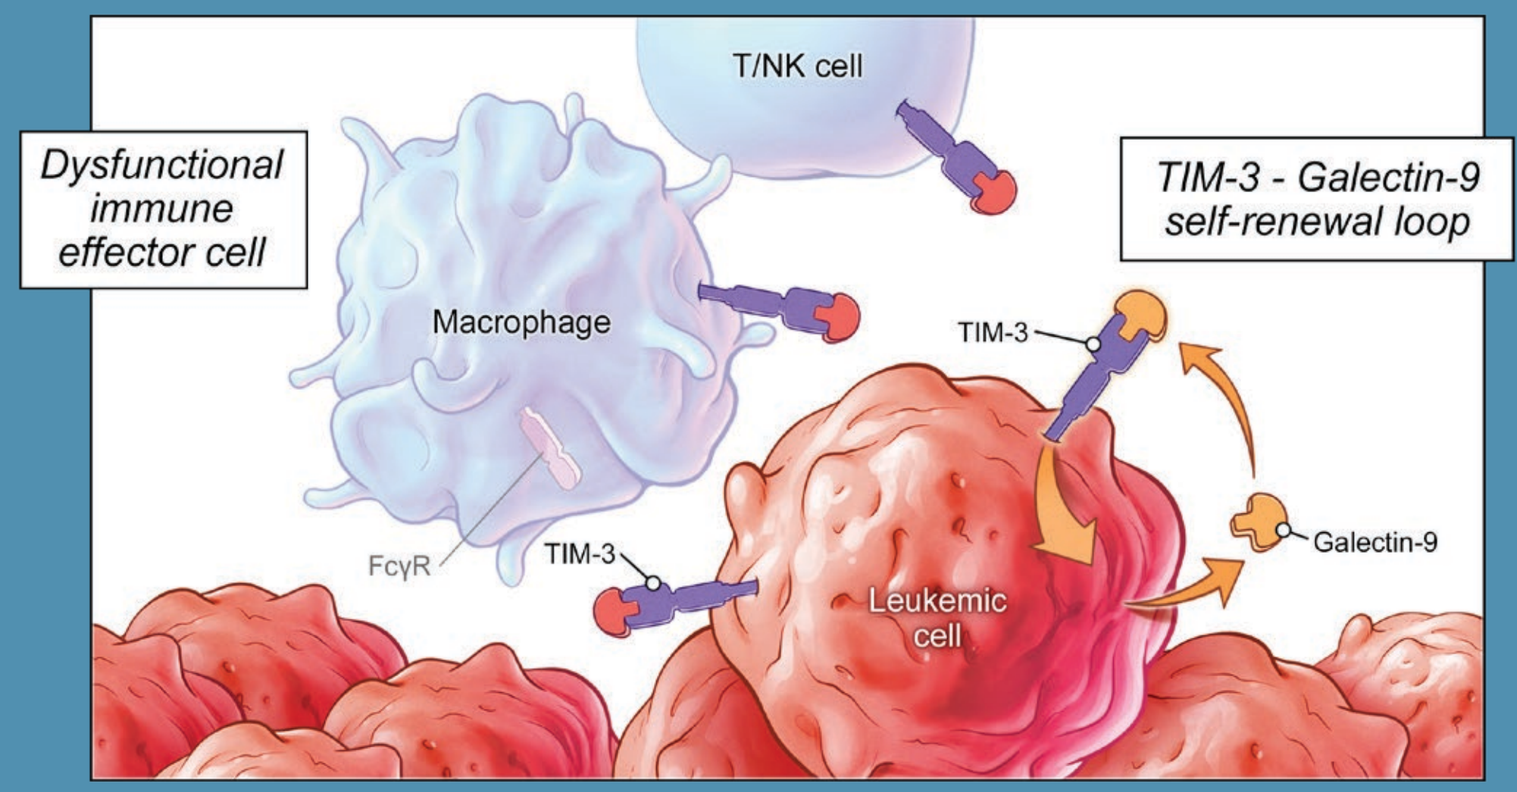
Dysfunctional immune effector cell
T/NK cell
TIM-3 - Galectin-9 self-renewal loop
Macrophage
TIM-3
FcγR
TIM-3
Leukemic cell
Galectin-9

FcγR, Fc gamma receptor; NK, natural killer; TIM-3, T-cell immunoglobulin domain and mucin domain-3.

Figure 2. Sabatolimab Targets TIM-3 on Immune and Leukemic Cells: A Novel Immuno-Myeloid Therapy

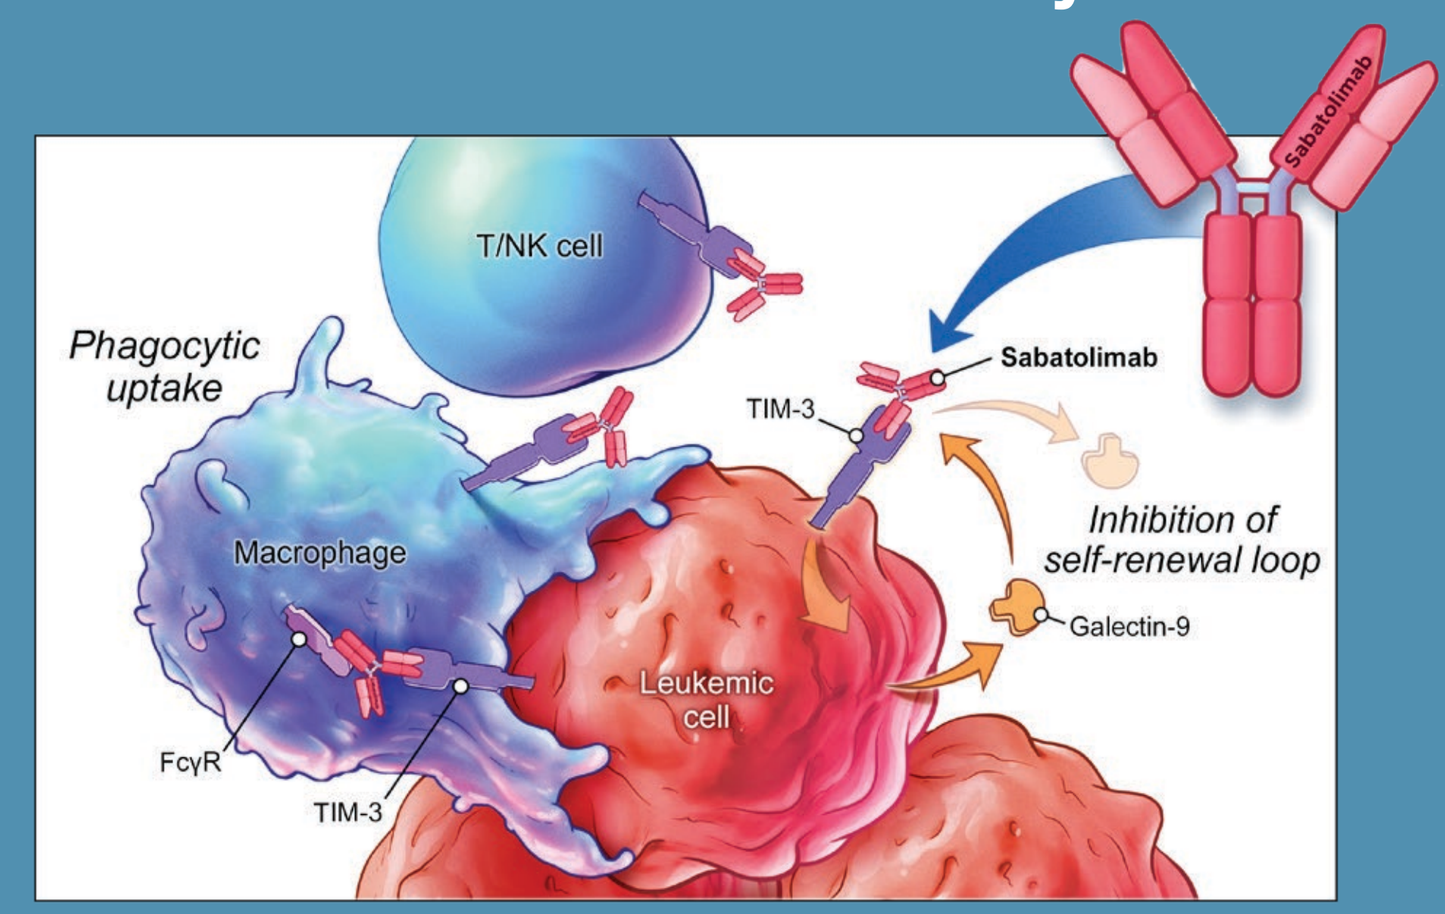
Sabatolimab
T/NK cell
Phagocytic uptake
Sabatolimab
TIM-3
Inhibition of self-renewal loop
Macrophage
Galectin-9
Leukemic cell
FcγR
TIM-3

FcγR, Fc gamma receptor; NK, natural killer; TIM-3, T-cell immunoglobulin domain and mucin domain-3.

METHODS

Figure 3. Trial Design: Phase Ib Study of Sabatolimab + HMA in MDS and AML

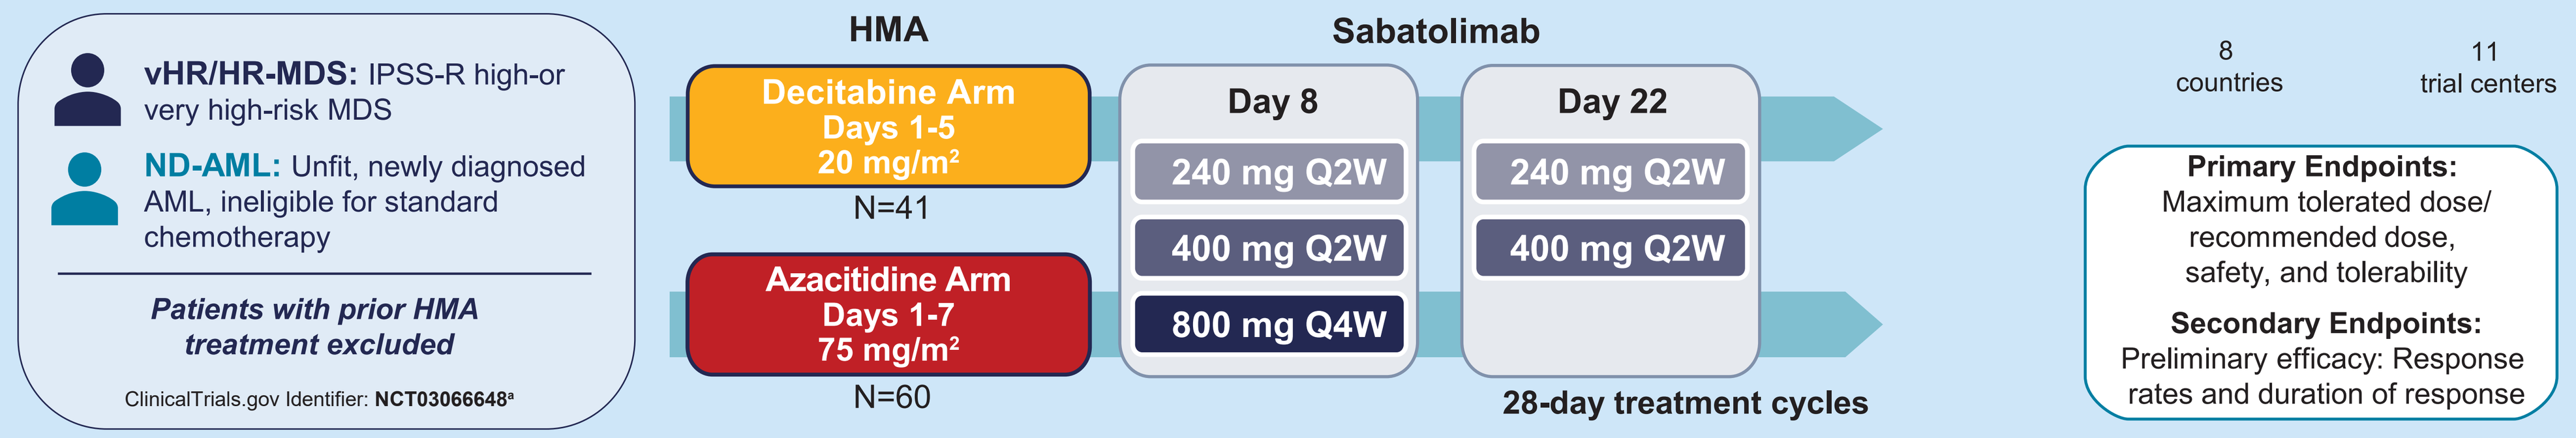
vHR/HR-MDS: IPSS-R high-or very high-risk MDS
ND-AML: Unfit, newly diagnosed AML, ineligible for standard chemotherapy
Patients with prior HMA treatment excluded
ClinicalTrials.gov Identifier: NCT03066648[a]
HMA
Decitabine Arm
Days 1-5
20 mg/m²
N=41
Azacitidine Arm
Days 1-7
75 mg/m²
N=60
Sabatolimab
Day 8
240 mg Q2W
400 mg Q2W
800 mg Q4W
Day 22
240 mg Q2W
400 mg Q2W
28-day treatment cycles
8 countries
11 trial centers
Primary Endpoints:
Maximum tolerated dose/ recommended dose, safety, and tolerability
Secondary Endpoints:
Preliminary efficacy: Response rates and duration of response

AML, acute myeloid leukemia; HMA, hypomethylating agent; HR, high-risk; IPSS-R, Revised International Prognostic Scoring System; MDS, myelodysplastic syndrome; ND, newly diagnosed; Q2W, every 2 weeks; Q4W, every 4 weeks; vHR, very high-risk.
[a]Multi-arm, open-label, Phase Ib dose-escalation and dose-expansion study of sabatolimab as a single-agent or in combination with HMAs or spartalizumab.

RESULTS

Table 1. Patient Characteristics

Parameter	vHR/HR-MDS n=53		ND-AML n=48	
Sabatolimab + decitabine, n	19		22	
Sabatolimab + azacitidine, n	34		26	
Median age (range), years	70.0 (23-90)		75.0 (59-89)	
Male, n (%)	29 (54.7)		26 (54.2)	
ECOG performance status, n (%)				
0	19 (35.8)		14 (29.2)	
1	29 (54.7)		29 (60.4)	
2	5 (9.4)		5 (10.4)	
Risk Category n (%)	IPSS-R[13]		2017 ELN risk[14]	
	High:	32 (60.4)	Intermediate:	19 (39.6)
	Very high:	21 (39.6)	Adverse:	29 (60.4)

AML, acute myeloid leukemia; ECOG, Eastern Cooperative Oncology Group; ELN, European LeukemiaNet; HR, high-risk; IPSS-R, Revised International Prognostic Scoring System; MDS, myelodysplastic syndrome; ND, newly diagnosed; vHR, very high-risk.

Table 2. Patient Disposition

		vHR/HR-MDS n=53	ND-AML n=48
Median exposure (range), mo	Sabatolimab + decitabine[a]	8.02 (0.9-33.5)	6.8 (0.8-30.9)
	Sabatolimab + azacitidine[b]	3.84 (0.8-17.2)	5.98 (1.1-16.7)
Ongoing,[c] n (%)		14 (26)	6 (12.5)
Discontinued, n (%)		39 (74)	42 (87.5)
Reason for discontinuation			
SCT		12 (23)	0
Disease progression		14 (26)	26 (54)
AE			
Unrelated to study treatment		0	2 (4)
Related to study treatment		0	1 (2)
Death			
Unrelated to study treatment		0	4 (8)
Related to study treatment		1[d] (2)	0
Patient decision		4 (8)	2 (4)
Physician decision		8 (15)	7 (15)
DLT		0	1 (3)[e]

AE, adverse event; AML, acute myeloid leukemia; ALT, alanine aminotransferase; DLT, dose limiting toxicity; HR, high-risk; MDS, myelodysplastic sydrome; ND, newly diagnosed; SCT, stem cell transplant; vHR, very high-risk.
[a]Enrollment started August 2017. [b]Enrollment started February 2019. [c]As of the cutoff date of April 1, 2021. [d]1 patient with neutropenic colitis reported as suspected to be related to study treatment died of septic shock. [e]Single DLT was a grade 3 event of elevated ALT/hepatitis.

Most AEs Consistent with HMA Alone

- Sabatolimab + HMA was safe and well tolerated in patients with vHR/HR-MDS and ND-AML (**Figure 4**)
- Patients infrequently required sabatolimab dose reduction:
 - 35/101 (35%) pts had dose interruption (cycle delay >7 days) due to adverse events (AEs)
 - 1/101 (1%) pts had dose reduction

Very Few Clinically Significant Treatment-Related imAEs

- 4/101 (4%) experienced a treatment-related possible imAE leading to sabatolimab discontinuation/reduction/interruption
- 4 pts with AML experienced grade 3 treatment-related possible immune-mediated AEs (imAEs)
- No grade 3 treatment-related possible imAEs in MDS
- No grade 4/5 treatment-related possible imAEs

Figure 4. Most Commonly Occurring AEs (≥15% in either population, regardless of relationship to treatment)

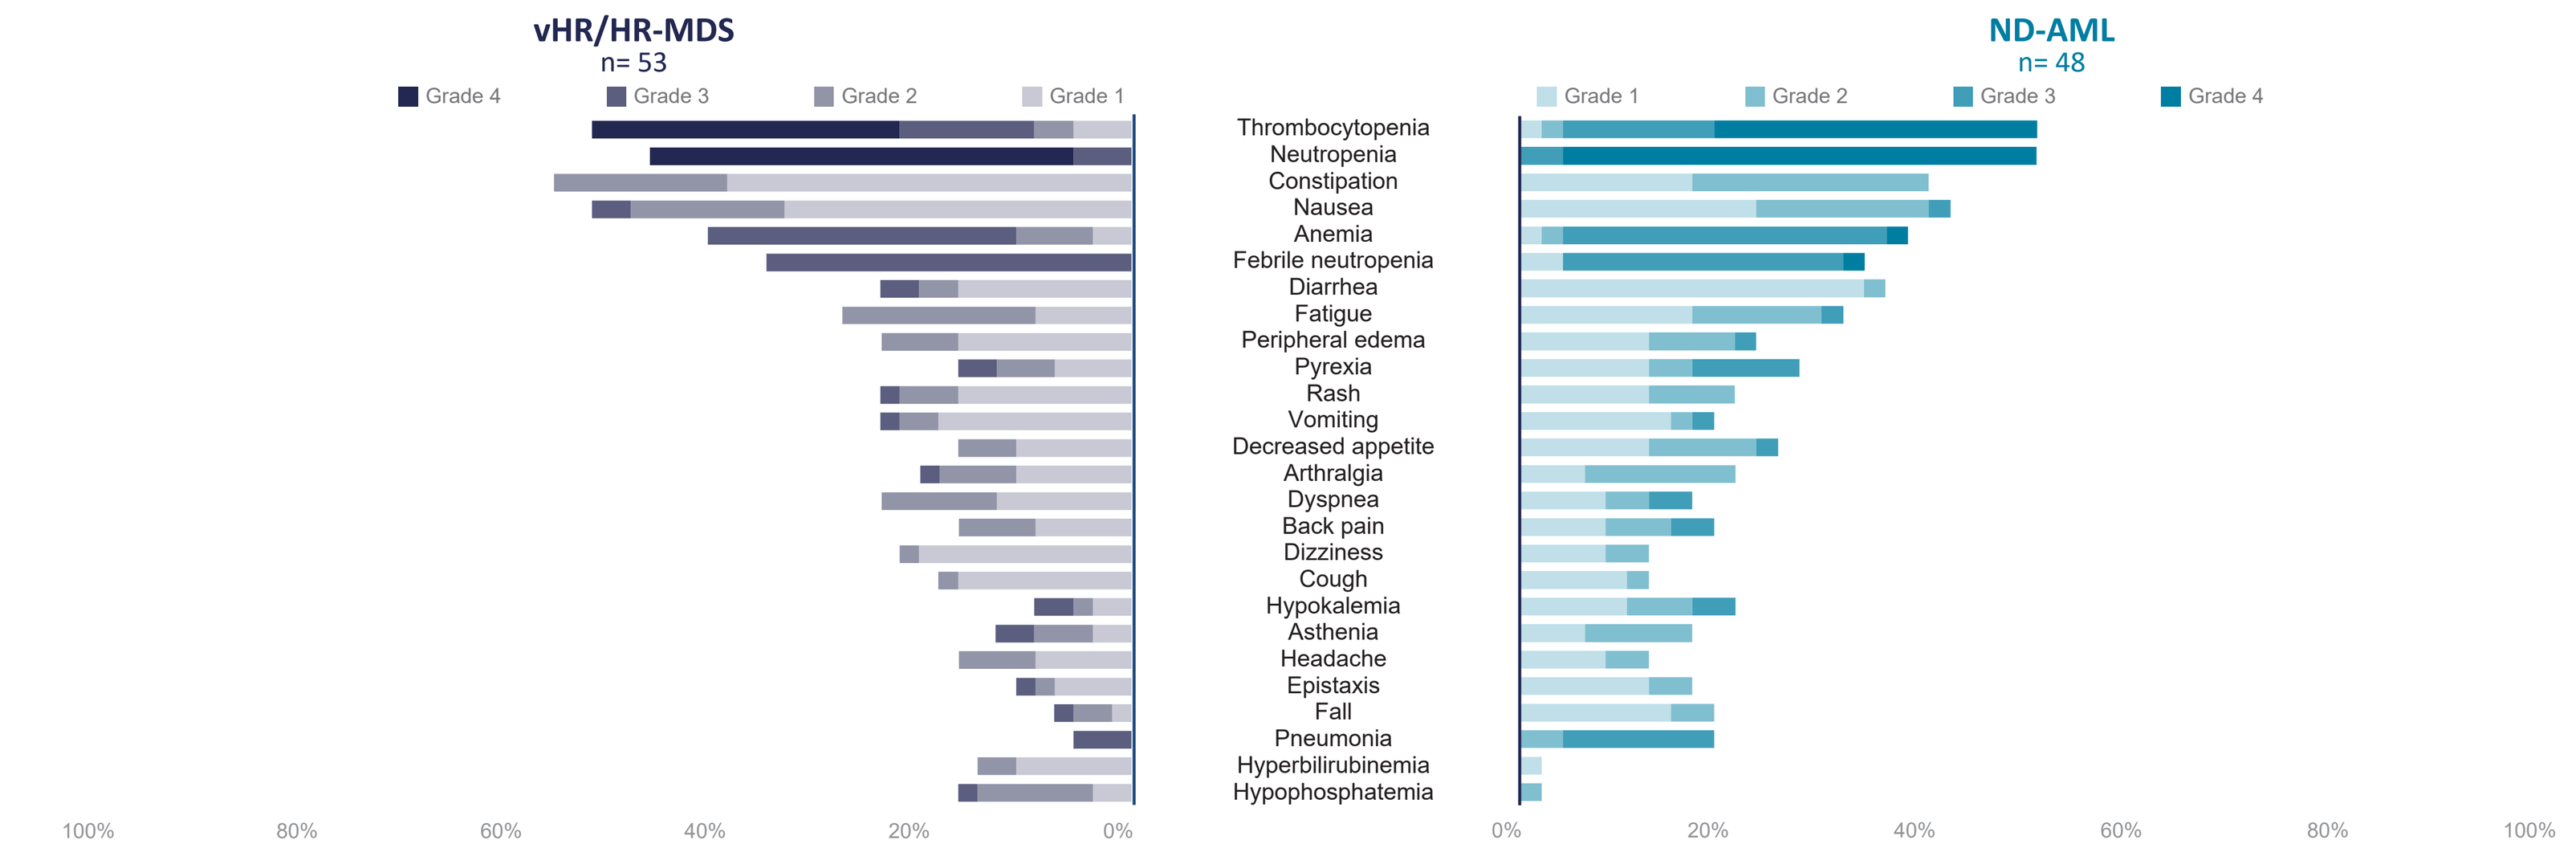
vHR/HR-MDS
n= 53
Grade 4
Grade 3
Grade 2
Grade 1
ND-AML
n= 48
Grade 1
Grade 2
Grade 3
Grade 4
Thrombocytopenia
Neutropenia
Constipation
Nausea
Anemia
Febrile neutropenia
Diarrhea
Fatigue
Peripheral edema
Pyrexia
Rash
Vomiting
Decreased appetite
Arthralgia
Dyspnea
Back pain
Dizziness
Cough
Hypokalemia
Asthenia
Headache
Epistaxis
Fall
Pneumonia
Hyperbilirubinemia
Hypophosphatemia
100%
80%
60%
40%
20%
0%
0%
20%
40%
60%
80%
100%

AE, adverse event; AML, acute myeloid leukemia; HR, high-risk; MDS, myelodysplastic syndrome; ND, newly diagnosed; vHR, very high-risk.

- Sabatolimab + hypomethylating agents (HMA) demonstrates promising response rates with emerging durability in very high-risk (vHR)/high-risk (HR)-MDS (**Figure 5**)

Figure 5. Response Rates and Durability Assessments in vHR/HR-MDS

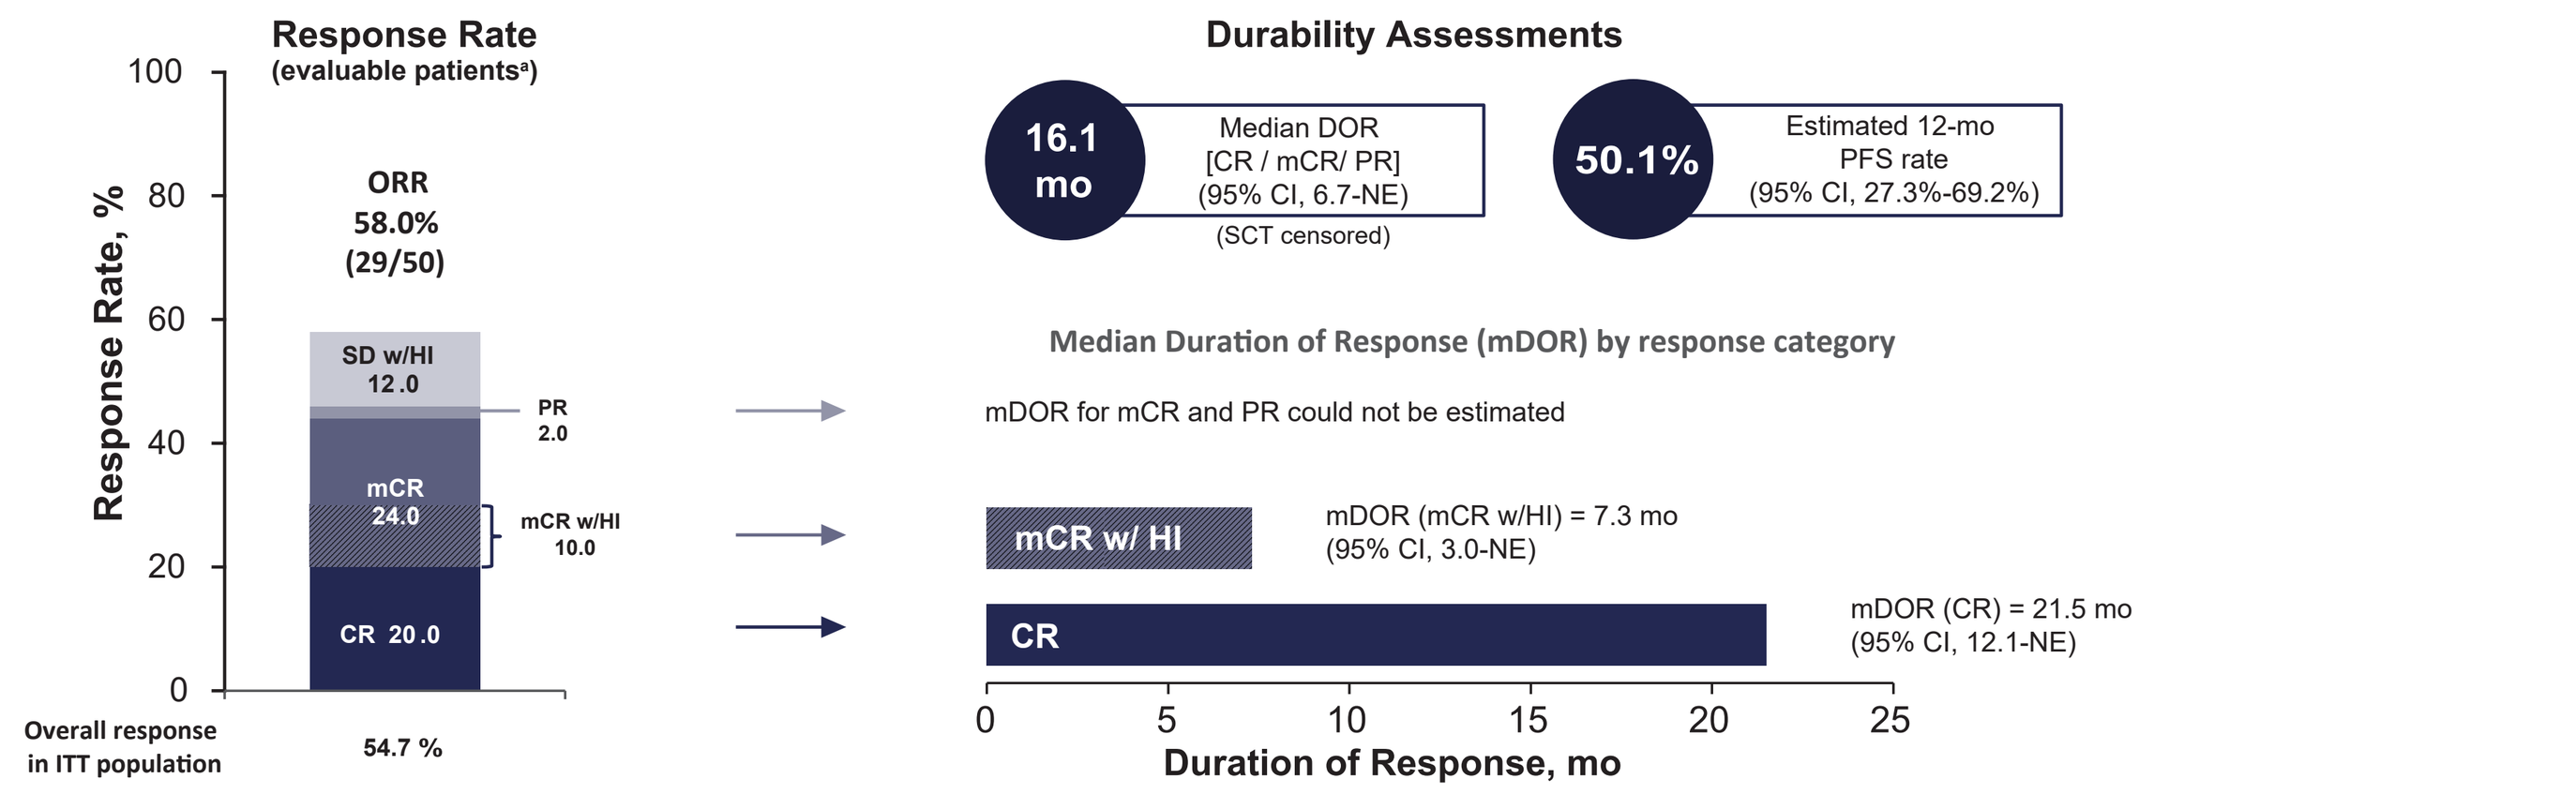
Response Rate
(evaluable patients[a])
Response Rate, %
ORR
58.0%
(29/50)
SD w/HI 12.0
PR 2.0
mCR 24.0
mCR w/HI 10.0
CR 20.0
Overall response in ITT population
54.7 %
Durability Assessments
16.1 mo
Median DOR [CR / mCR/ PR] (95% CI, 6.7-NE)
(SCT censored)
50.1%
Estimated 12-mo PFS rate (95% CI, 27.3%-69.2%)
Median Duration of Response (mDOR) by response category
mDOR for mCR and PR could not be estimated
mCR w/ HI
mDOR (mCR w/HI) = 7.3 mo (95% CI, 3.0-NE)
CR
mDOR (CR) = 21.5 mo (95% CI, 12.1-NE)
Duration of Response, mo

CI, confidence interval; CR, complete remission; DOR, duration of response; HI, hematologic improvement; HR, high-risk; ITT, intent-to-treat; mCR, bone marrow complete remission; mDOR, median duration of response; MDS, myelodysplastic syndrome; NE, not estimated; ORR, overall response rate; PFS, progression-free survival; PR, partial remission; SCT, stem cell transplant; SD, stable disease; vHR, very high-risk.
[a]Evaluable patients, including patients with a valid baseline and at least 1 postbaseline bone marrow assessment or if they had disease progression or disease-related death prior to the first marrow assessment.

- Sabatolimab + HMA demonstrates promising response rates with emerging durability in newy diagnosed (ND)-AML (**Figure 6**)

Figure 6. Response Rates and Durability Assessments in ND-AML

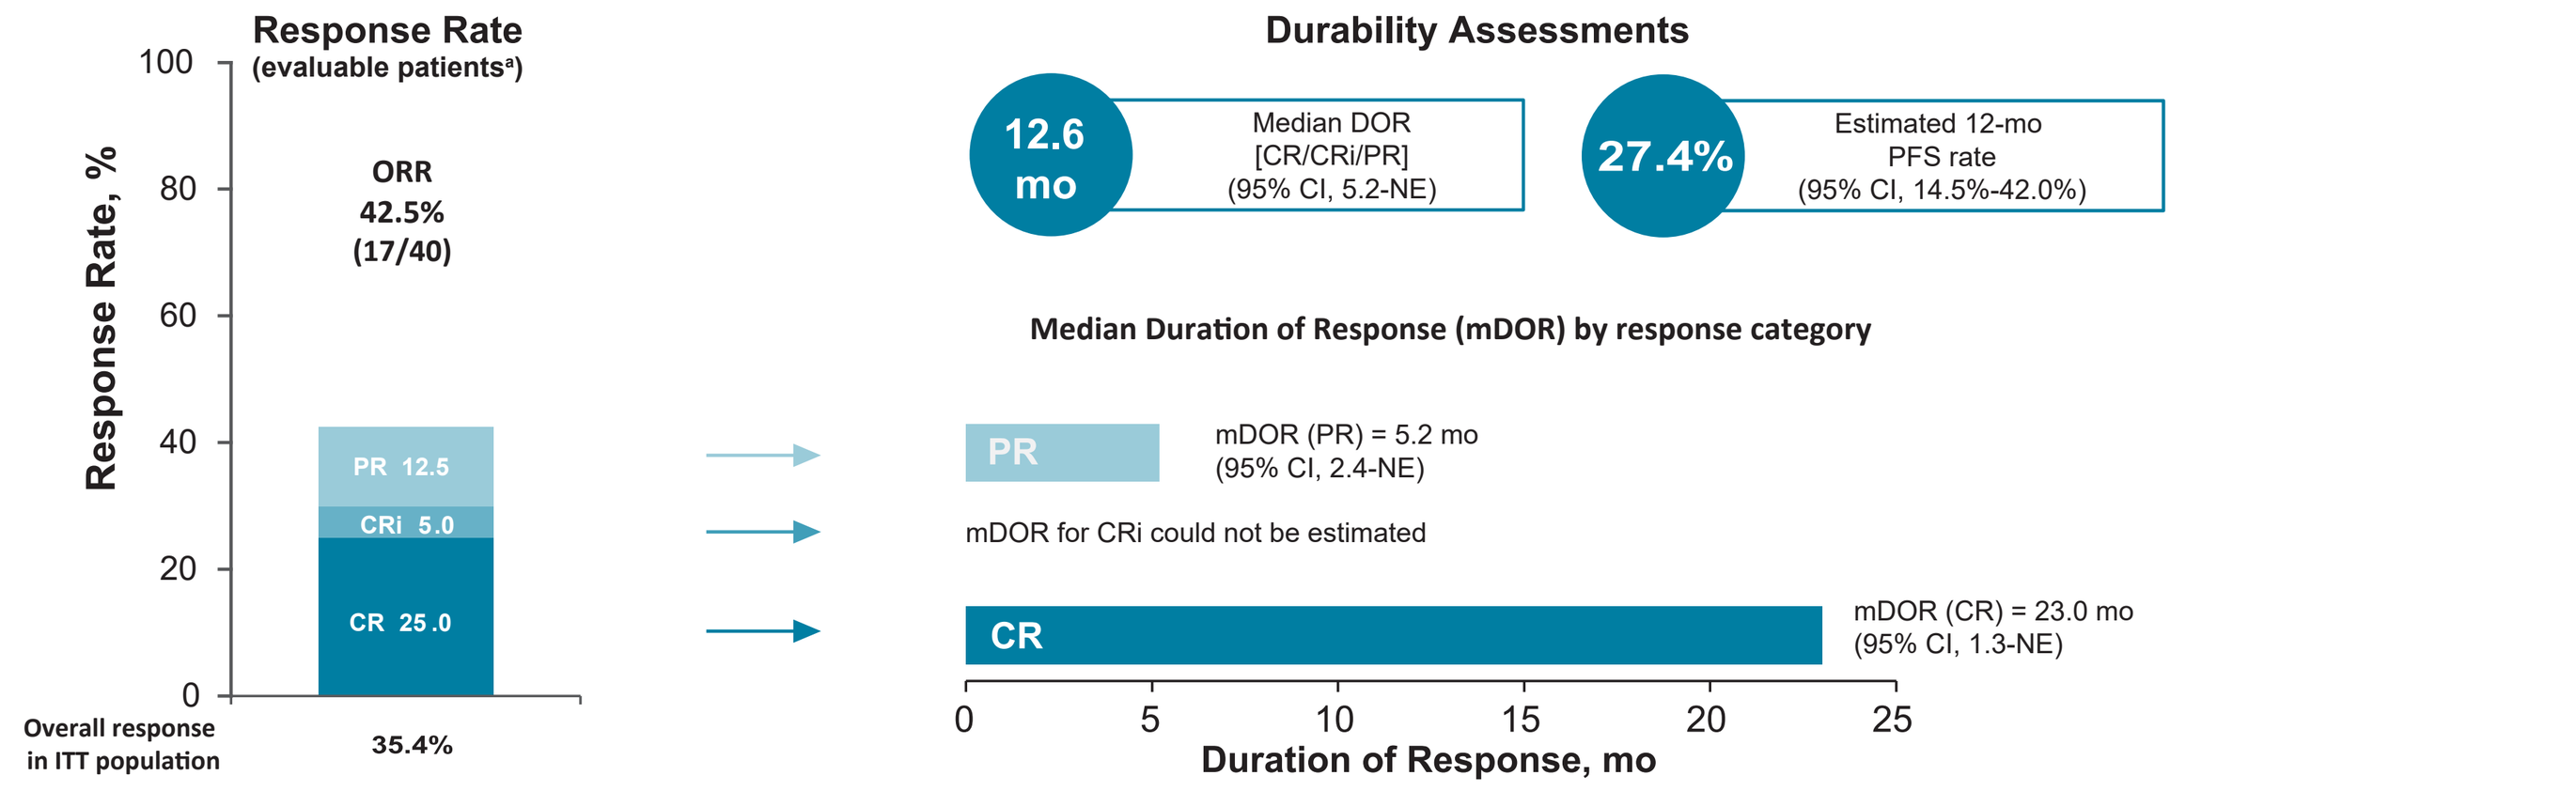
Response Rate
(evaluable patients[a])
Response Rate, %
ORR
42.5%
(17/40)
PR 12.5
CRi 5.0
CR 25.0
Overall response in ITT population
35.4%
Durability Assessments
12.6 mo
Median DOR [CR/CRi/PR] (95% CI, 5.2-NE)
27.4%
Estimated 12-mo PFS rate (95% CI, 14.5%-42.0%)
Median Duration of Response (mDOR) by response category
PR
mDOR (PR) = 5.2 mo (95% CI, 2.4-NE)
mDOR for CRi could not be estimated
CR
mDOR (CR) = 23.0 mo (95% CI, 1.3-NE)
Duration of Response, mo

AML, acute myeloid leukemia; CI, confidence interval; CR, complete remission; CRi, complete remission with incomplete count recovery; DOR, duration of response; ITT, intent-to-treat; mDOR, median duration of response; ND, newly diagnosed; NE, not estimated; ORR, objective response rate; PFS, progression-free survival; PR, partial remission.
[a]Evaluable patients, including patients with a valid baseline and at least 1 postbaseline bone marrow assessment or if they had disease progression or disease-related death prior to the first marrow assessment.

Sabatolimab + HMA Differentially Regulate IL-1β in Immune vs Leukemic Cells

- Interleukin-1β (IL-1β) is a pleiotropic cytokine with key roles in inflammation and MDS/AML
 - IL-1β signaling has been shown to promote leukemic cell growth[15] and blockade blunts AML and LSC growth[16,17]
 - Patients with germline TIM-3 inactivation have enhanced myeloid cell secretion of IL-1β[18]
- IL-1β is the most differentially expressed gene in the bone marrow of sabatolimab + HMA responders vs. non-responders (**Figure 7**), with differential regulation in myeloid immune vs leukemic cells (**Figure 8**)

Figure 7. RNA-seq: IL-1β is Downregulated in Patients Responding to Sabatolimab + HMA

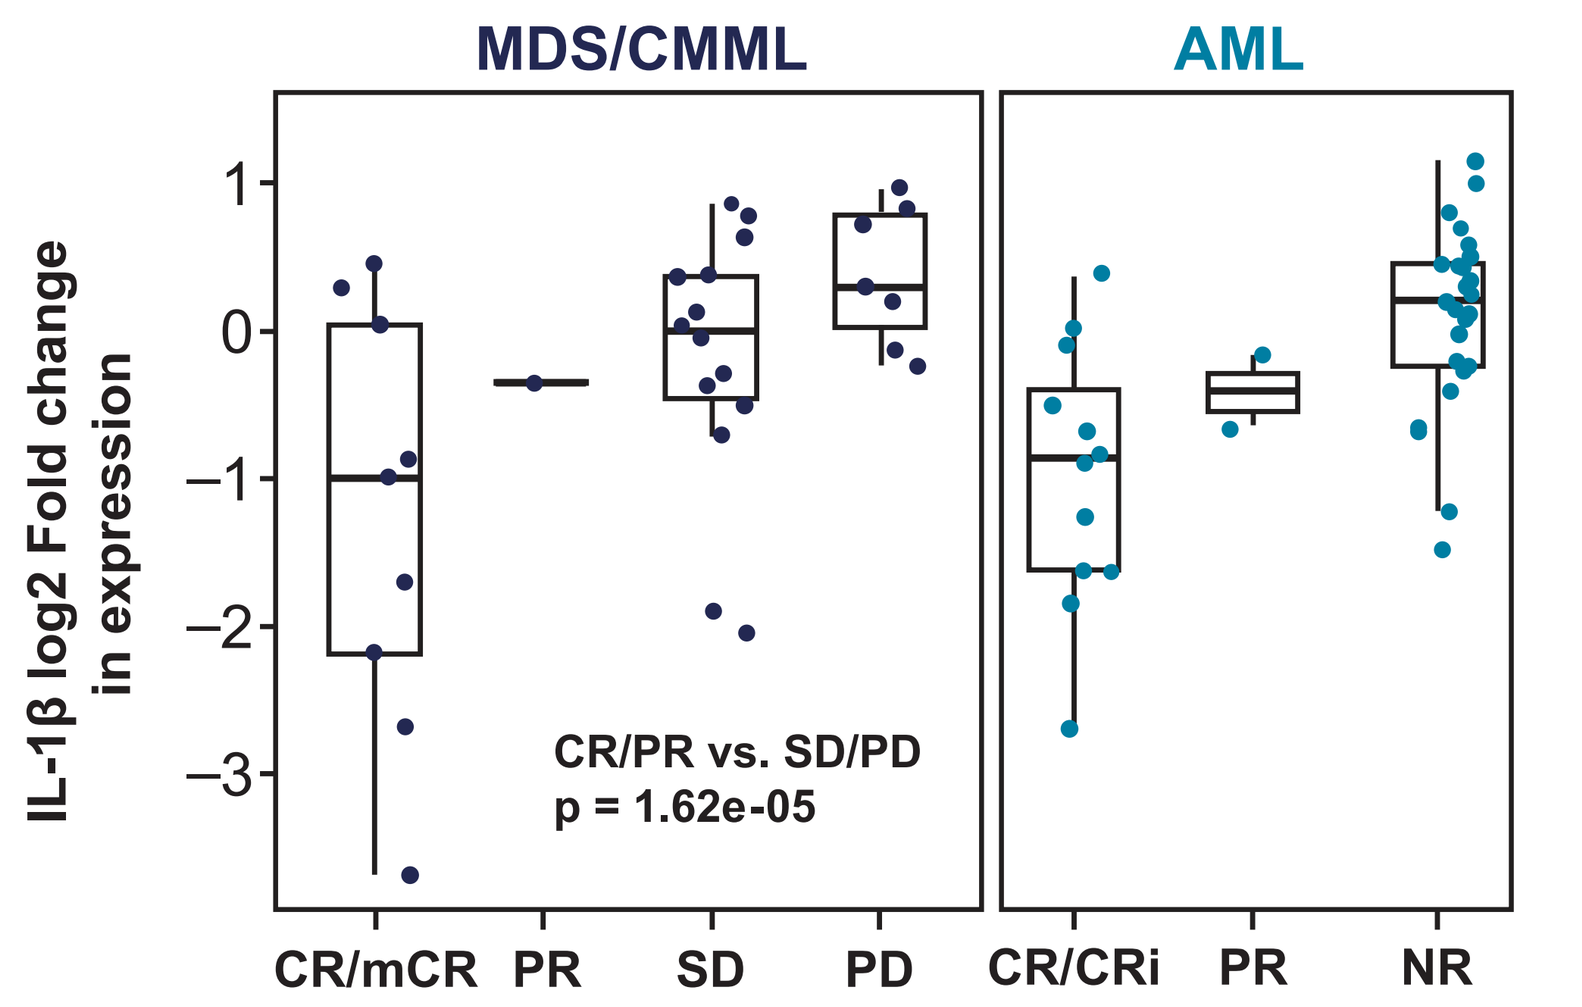
MDS/CMML
AML
IL-1β log2 Fold change in expression
CR/PR vs. SD/PD
p = 1.62e-05
CR/mCR
PR
SD
PD
CR/CRi
PR
NR

Figure 8. scRNA-seq: IL-1β is Downregulated in Blasts & LSCs, but Increased in Myeloid Cells Post-Sabatolimab + HMA

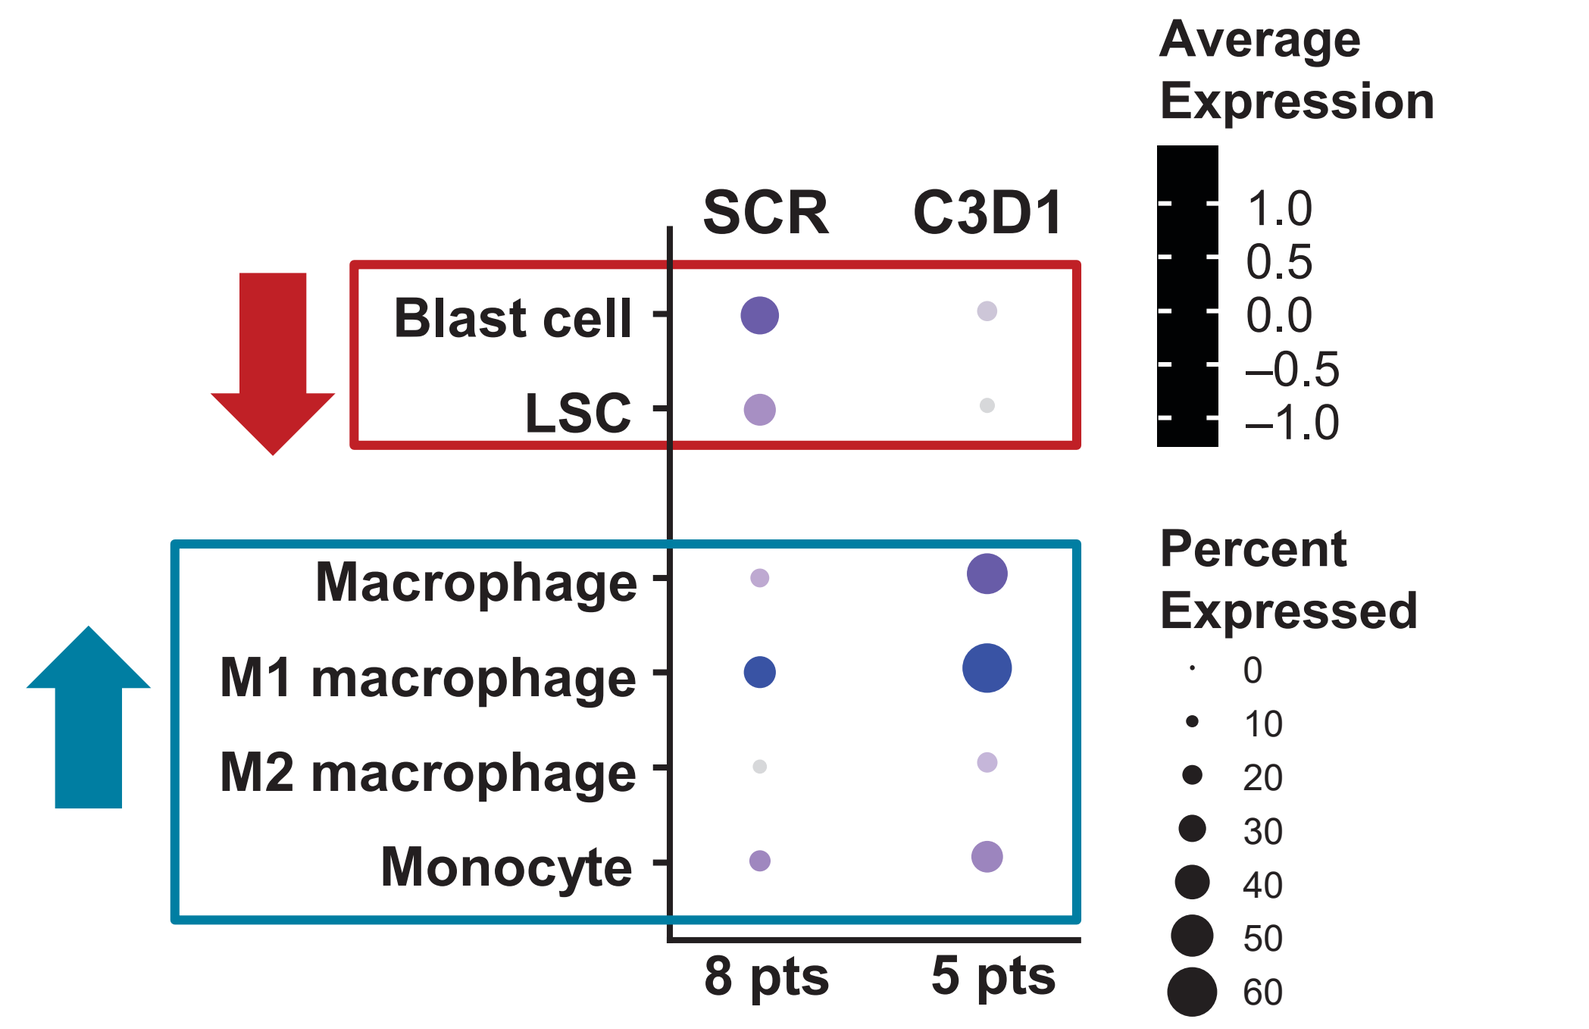
SCR
C3D1
Blast cell
LSC
Macrophage
M1 macrophage
M2 macrophage
Monocyte
8 pts
5 pts
Average Expression
1.0
0.5
0.0
−0.5
−1.0
Percent Expressed
0
10
20
30
40
50
60

AML, acute myeloid leukemia; C, cycle; CMML, chronic myelomonocytic leukemia; CR, complete remission; CRi, complete remission with incomplete count recovery; D, day; HMA, hypomethylating agent; IL-1β, interleukin 1-beta; LSC, leukemic stem cell; mCR, bone marrow complete remission; MDS, myelodysplastic syndrome; NR, no response; PD, progressive disease; PR, partial remission; RNA-seq, RNA sequencing; scRNA-seq, single-cell RNA sequencing; SCR, screening; SD, stable disease.

- Patients with adverse risk disease were able to achieve durable responses (**Figure 9**)

Figure 9. Response Rates in vHR/HR-MDS and ND-AML

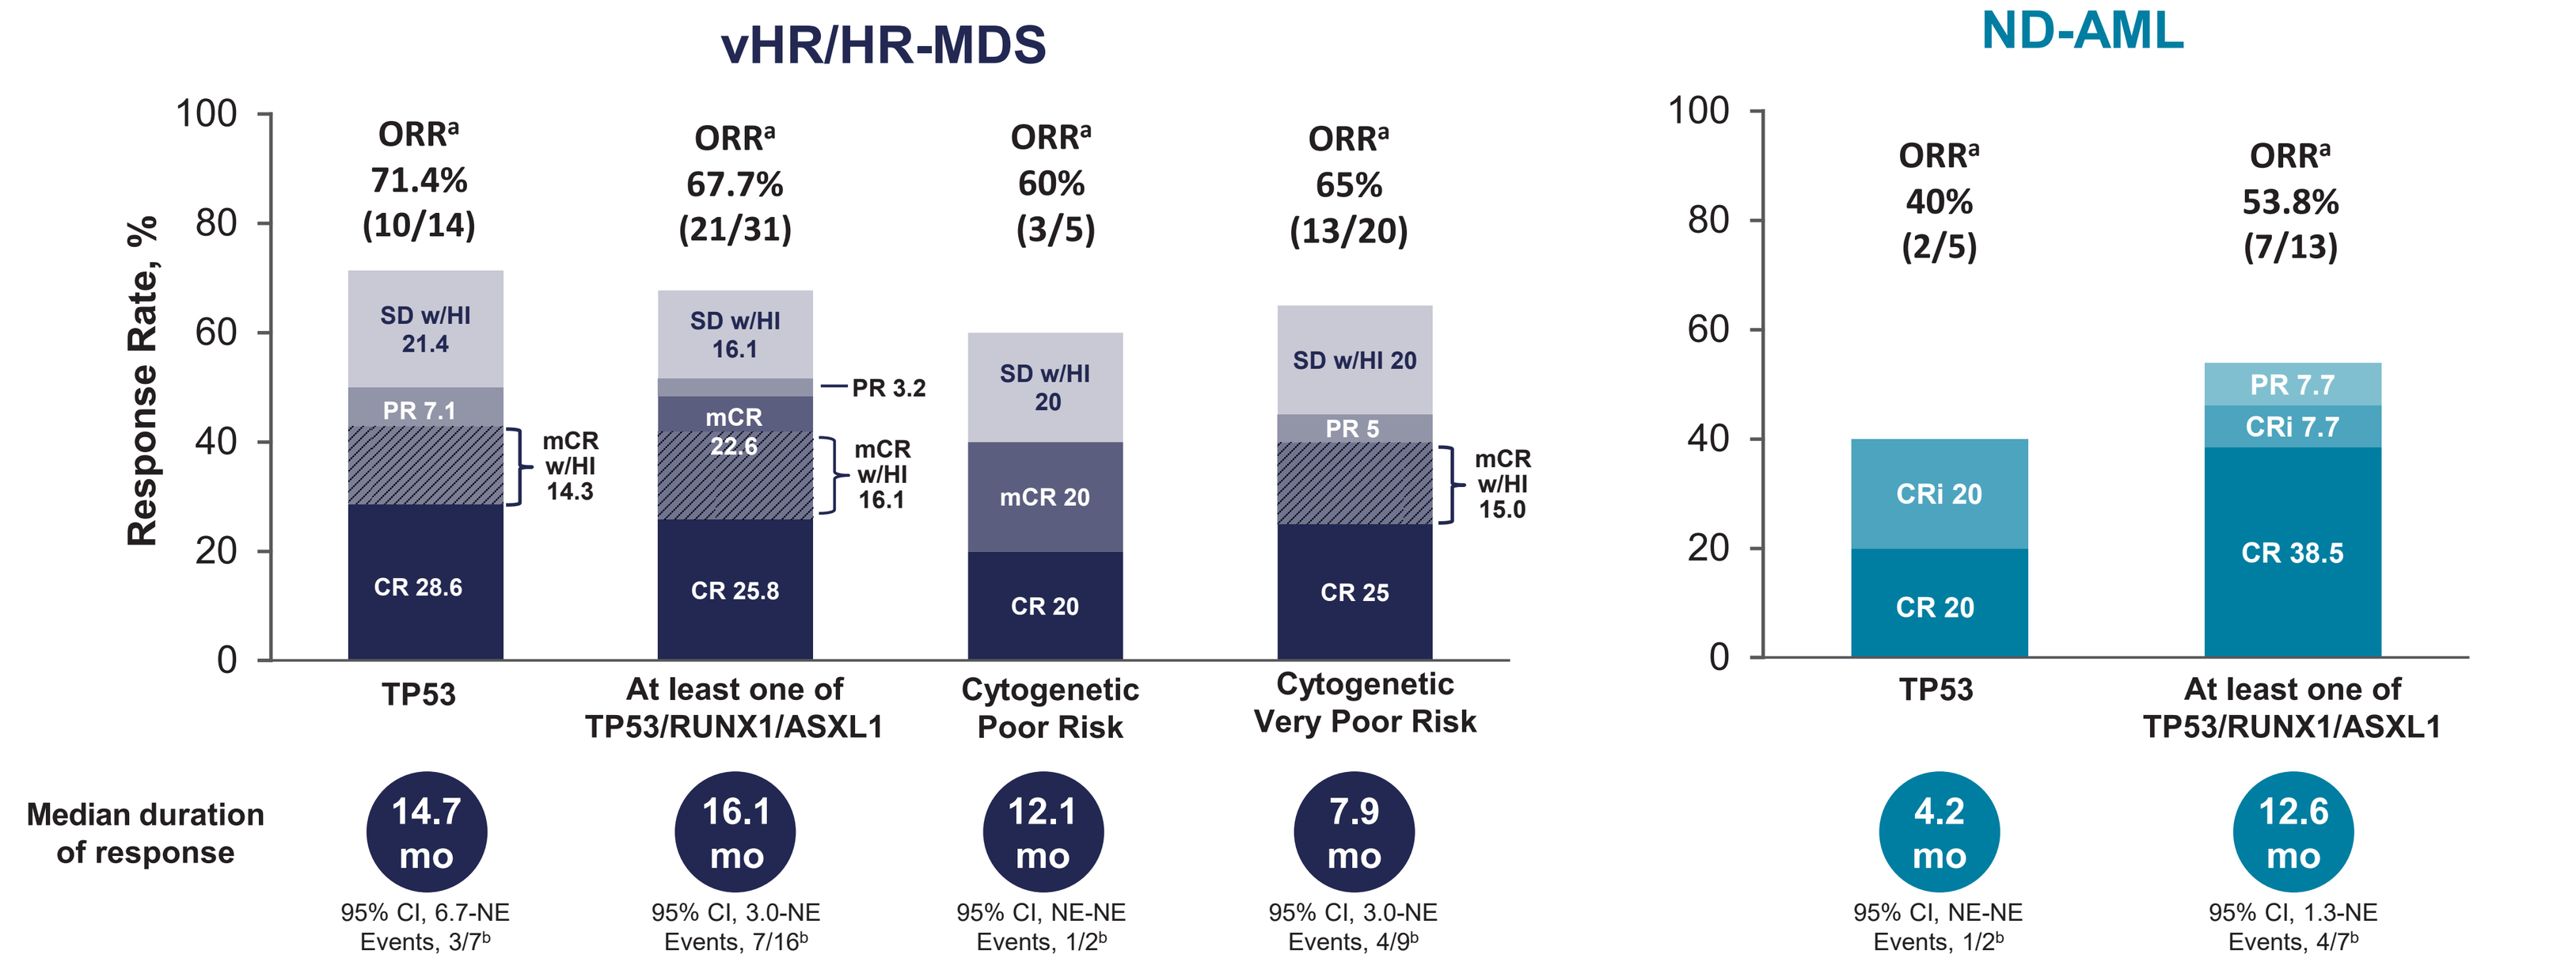
vHR/HR-MDS
ND-AML
Response Rate, %
ORR[a] 71.4% (10/14)
ORR[a] 67.7% (21/31)
ORR[a] 60% (3/5)
ORR[a] 65% (13/20)
ORR[a] 40% (2/5)
ORR[a] 53.8% (7/13)
SD w/HI 21.4
PR 7.1
mCR w/HI 14.3
CR 28.6
SD w/HI 16.1
PR 3.2
mCR 22.6
mCR w/HI 16.1
CR 25.8
SD w/HI 20
mCR 20
CR 20
SD w/HI 20
PR 5
mCR w/HI 15.0
CR 25
CRi 20
CR 20
PR 7.7
CRi 7.7
CR 38.5
TP53
At least one of TP53/RUNX1/ASXL1
Cytogenetic Poor Risk
Cytogenetic Very Poor Risk
TP53
At least one of TP53/RUNX1/ASXL1
Median duration of response
14.7 mo
95% CI, 6.7-NE
Events, 3/7[b]
16.1 mo
95% CI, 3.0-NE
Events, 7/16[b]
12.1 mo
95% CI, NE-NE
Events, 1/2[b]
7.9 mo
95% CI, 3.0-NE
Events, 4/9[b]
4.2 mo
95% CI, NE-NE
Events, 1/2[b]
12.6 mo
95% CI, 1.3-NE
Events, 4/7[b]

AML, acute myeloid leukemia; BOR, best overall response; CI, confidence interval; CR, complete remission; CRi, complete remission with incomplete count recovery; DOR, duration of response; HI, hematologic improvement; HR, high-risk; MDS, myelodysplastic syndrome; mCR, bone marrow complete remission; ND, newly diagnosed; NE, not estimated; ORR, objective response rate; SD, stable disease; PR, partial remission; vHR, very high-risk.
[a]ORR for patients with MDS was defined as CR + mCR + PR + SD with HI; ORR for patients with ND-AML was defined as CR + CRi + PR. [b]DOR events (including progression/relapse and death) reported out of the number of patients with a BOR of CR, mCR, or PR (for MDS) or CR, CRi, or PR (for AML).

Acknowledgements

The authors thank the patients who participated in the trial and their families.

Financial support for medical editorial assistance was provided by Novartis Pharmaceuticals Corporation. The authors thank Kendall Foote, PhD, of Medical Expressions, for medical editorial assistance with this poster presentation.

Disclosures

GG-M reports consultancy for Acceleron Pharmaceuticals, Astex Pharmaceuticals, Bristol-Myers Squibb, Celgene, Helsinn Therapeutics, Jazz Pharmaceuticals, and Genentech; honoraria for AbbVie, Acceleron Pharmaceuticals, Astex Pharmaceuticals, Celgene, and Helsinn Therapeutics; research funding from AbbVie, Amphivena Therapeutics, Astex Pharmaceuticals, Bristol-Myers Squibb, Celgene, Helsinn Therapeutics, H3 Biomedicine, Merck, Novartis, Genentech, and Onconova; membership on an entity's Board of Directors or advisory committees for Genentech. **AHW** reports consultancy for Servier, BMS; honoraria for AbbVie, Servier, Novartis, AstraZeneca, Pfizer, BMS, Janssen, Amgen, Astellas, Genentech, and Celgene; research funding from Abbvie, Servier, Novartis, AstraZeneca, BMS, Amgen, and Celgene; speakers bureau for AbbVie, BMS, and Novartis; patents and royalties from Walter and Eliza Hall Institute of Medical Research; membership on an entity's Board of Directors or advisory committees for Astellas, Genentech, and Celgene. **KP** reports consultancy for Novartis; honoraria and research funding from Novartis and BMS/Celgene. **SK** reports honoraria, membership on an entity's Board of Directors or advisory committee and research funding for Novartis. **ET** reports consultancy for AbbVie, and Notable Labs; research funding for Incyte; membership on an entity's Board of Directors or advisory committees for Agios, Genentech, AbbVie, Astellas, and Daiichi Sankyo; current equity holder in private company with Notable Labs. **MW** reports honoraria for MacroGenics. **JJ** reports honoraria for Novartis, Pfizer, and Incyte; research funding from Novartis, and BMS, membership on an entity's Board of Directors or advisory committees for Novartis, Pfizer, AbbVie, and Incyte; Founder of the HematologyApp which is supported by Janssen, BMS Incyte, MDS, Pfizer, Daiichi-Sankyo, Roche and Takeda. **RN** reports current spouse employment with Sanofi-Genzyme; prior spouse employment within 24 months with Takeda; prior spouse employment within 24 months and prior spouse equity divested within past 24 months with Genentech. **MK** reports consultancy with Astellas; research funding with AbbVie; membership on an entity's Board of Directors or advisory committees with Jazz Pharma, Astellas, Novartis, and Celgene; speakers bureau with Novartis, and Celgene. **OO** reports honoraria for Novartis, Amgen, Celgene, Fusion Pharma, and Incyte; research funding from Amgen, Celgene, and Incyte. **MLR** reports employment at Novartis and consultancy for Qiagen. **UB** reports research funding from Takeda, Novartis, Jazz Pharmaceuticals, and Pfizer; membership on an entity's Board of Directors or advisory committees for Genentech, Daiichi Sankyo, Takeda, Novartis, and Pfizer; investigator in AbbVie-funded clinical trials. **AMB** reports consultancy for Acceleron Pharma Inc., Biogen, Celgene/BMS, Forty Seven, Inc., Jazz Pharma, Novartis, Takeda, and Xcenda, research funding from Celgene/BMS, Novartis, Takeda, GSK, Janssen, and AstraZeneca. **PN, EO, NP, JM, FM, NZ,** and **AM** are employees at Novartis**.** All other authors had nothing to disclose.

References

1. Pardoll DM. *Nat Rev Cancer*. 2012;12(4):252-264. **2.** Das M, et al. *Immunol Rev*. 2017;276(1):97-111. **3.** Kikushige Y, Miyamoto T. *Int J Hematol*. 2013;98(6):627-633. **4.** Kikushige Y, et al. *Cell Stem Cell*. 2010;7(6):708-717. **5.** Ngiow SF. *Cancer Res*. 2011;71(10):3540-3551. **6.** Sakuishi K, et al. *Trends Immunol*. 2011;32(8):345-349. **7.** Sabatos-Peyton C. *AACR 2016*. Oral presentation. **8.** Borate U, et al. *ASH 2019*. Oral presentation. **9.** Acharya N, et al. *J Immunother Cancer*. 2020;8(1):e000911. **10.** Sabatos-Peyton C, et al. *SITC 2020*. Abstract 439. **11.** Borate U, et al. *HemaSphere*. 2020;4(suppl 1):Abstract S185. **12.** Borate U, et al. *EHA 2020*. Oral presentation. **13.** Greenberg PL, et al. *Blood*. 2012;120(12):2454-2465. **14.** Döhner H, et al. *Blood*. 2017;129(4):424-447. **15.** Carey A, et al. *Cell Reports*. 2017;18:3204. **16.** Estrov Z, et al. *Blood*. 1992;79:1938-1945. **17.** Cozzolino F, et al. *Proc Natl Acad Sci USA*. 1989;86:2369-2373. **18.** Gayden T, et al. *Nat Genet*. 2018;50:1650-1657.