MDS-404

Guillermo Garcia-Manero, ggarciam@mdanderson.org

Population Pharmacokinetics Modeling and Evaluation of Clinical Efficacy and Safety of Sabatolimab: Dose Selection and Dose-Response Analysis

Guillermo Garcia-Manero,[1] Andrew H. Wei,[2] Kimmo Porkka,[3] Steve Knapper,[4] Norbert Vey,[5] Sebastian Scholl,[6] Martin Wermke,[7] Jeroen Janssen,[8] Elie Traer,[9] Sun Loo,[10] Rupa Narayan,[11] Natalia Tovar,[12] Mika Kontro,[3] Oliver Ottmann,[4] Siyan Xu,[13] Andrew Lewandowski,[13] Andrew Stein,[13] Purushotham Naidu,[14] Na Zhang,[13] Anisa Mohammed,[14] Mikael L. Rinne,[13] Haiying Sun,[14] Andrew M. Brunner,[11] Uma Borate[9]

[1]MD Anderson Cancer Center, Houston, TX, USA; [2]The Alfred Hospital and Monash University, Melbourne, Victoria, Australia; [3]Helsinki University Hospital Comprehensive Cancer Center, Department of Hematology, Helsinki, Finland; [4]Cardiff University, Cardiff, UK; [5]Institut Paoli-Calmettes, Marseille, France; [6]University Hospital Jena, Jena, Germany; [7]University Hospital Dresden, Dresden, Germany; [8]Amsterdam University Medical Centers, VU University Medical Center, Amsterdam, The Netherlands; [9]Oregon Health & Science University, Portland, OR, USA; [10]The Alfred Hospital, Melbourne, Victoria, Australia; [11]Massachusetts General Hospital, Boston, MA, USA; [12]Hospital Clínic, Barcelona, Spain; [13]Novartis Institutes for BioMedical Research, Cambridge, MA, USA; [14]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA

Scan to obtain:
- Poster

http://novartis.medicalcongressposters.com/Default.aspx?doc=24005
Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

CONCLUSIONS

- Sabatolimab 400 mg Q2W was predicted to have the highest steady state C_{trough} and TIM-3 occupancy rate when combined with HMA; 800 mg was predicted to be an equivalent Q4W dosing regimen
- Sabatolimab 400 mg Q2W and 800 mg Q4W provided similarly high levels of TIM-3 engagement based on analysis of sTIM-3 concentration and receptor occupancy modeling
- Sabatolimab + HMA showed promising efficacy and good safety/tolerability in patients with vHR/HR-MDS and AML at both 400 mg Q2W and 800 mg Q4W, with no clear relationship between sabatolimab dose/exposure and safety/efficacy
- These results validate the selection of the 400 mg Q2W and 800 mg Q4W dosing regimens for the STIMULUS clinical trial program

This study is sponsored by Novartis Pharmaceuticals Corporation.
Presented at the Society of Hematologic Oncology Ninth Hybrid Annual Meeting; September 8-11, 2021.

INTRODUCTION

TIM-3

- T-cell immunoglobulin domain and mucin domain-3 (TIM-3) is an inhibitory receptor expressed on macrophages, monocytes, natural killer cells, dendritic cells, and T cells[1,2] (**Figure 1**)
- Involved in regulating innate and adaptive immune responses[1,2]
- Expressed on leukemic stem cells (LSCs)/blasts but not normal hematopoietic stem cells (HSCs),[3,4] making it a promising target in myelodysplastic syndrome (MDS) and acute myeloid leukemia (AML)[4-6]

Sabatolimab

- High-affinity, humanized, immunoglobulin G4 anti–TIM-3 monoclonal antibody[6,7]
- Binds TIM-3 on immune cells, enhancing antileukemic immune activation and phagocytic killing of LSCs and blasts[2,7-9] **(Figure 2)**
- Directly targets TIM-3 on LSCs, inhibiting TIM-3/galectin-9-driven self-renewal[2,9]

Figure 1. TIM-3 is an Inhibitory Receptor Expressed on Immune and Leukemic Cells

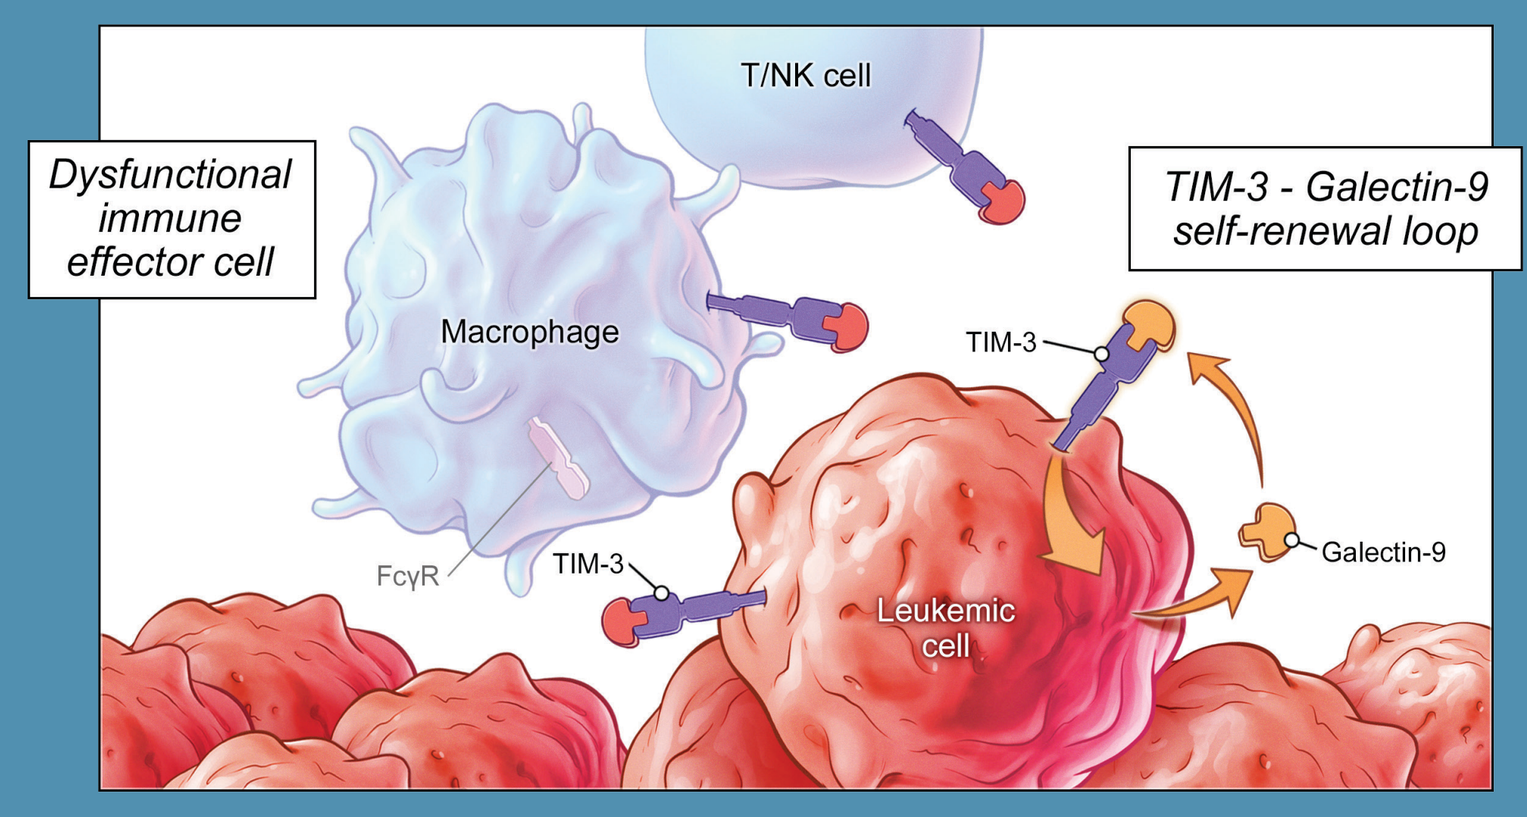
T/NK cell
Dysfunctional immune effector cell
TIM-3 - Galectin-9 self-renewal loop
Macrophage
TIM-3
FcγR
TIM-3
Leukemic cell
Galectin-9

FcγR, Fc gamma receptor; NK, natural killer; TIM-3, T-cell immunoglobulin domain and mucin domain-3.

Figure 2. Sabatolimab Targets TIM-3 on Immune and Leukemic Cells: A Novel Immuno-Myeloid Therapy

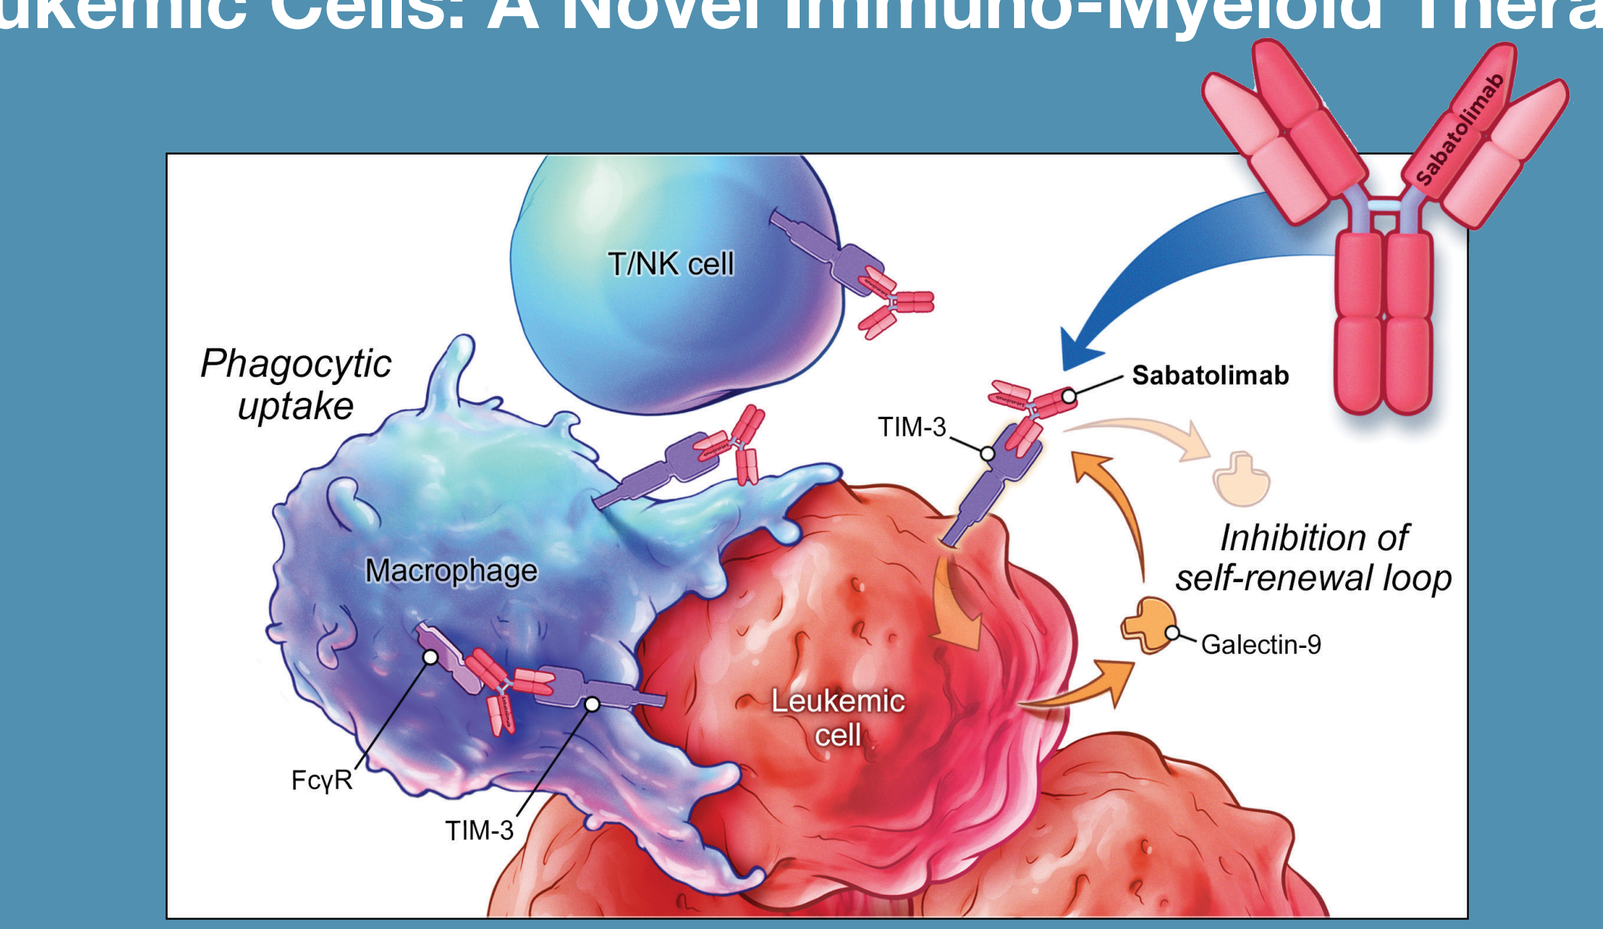
Sabatolimab
T/NK cell
Phagocytic uptake
Sabatolimab
TIM-3
Inhibition of self-renewal loop
Macrophage
Leukemic cell
Galectin-9
FcγR
TIM-3

FcγR, Fc gamma receptor; NK, natural killer; TIM-3, T-cell immunoglobulin domain and mucin domain-3.

METHODS

Figure 3. Evaluation of Sabatolimab Pharmacokinetics and Dose Response in Early Clinical Studies

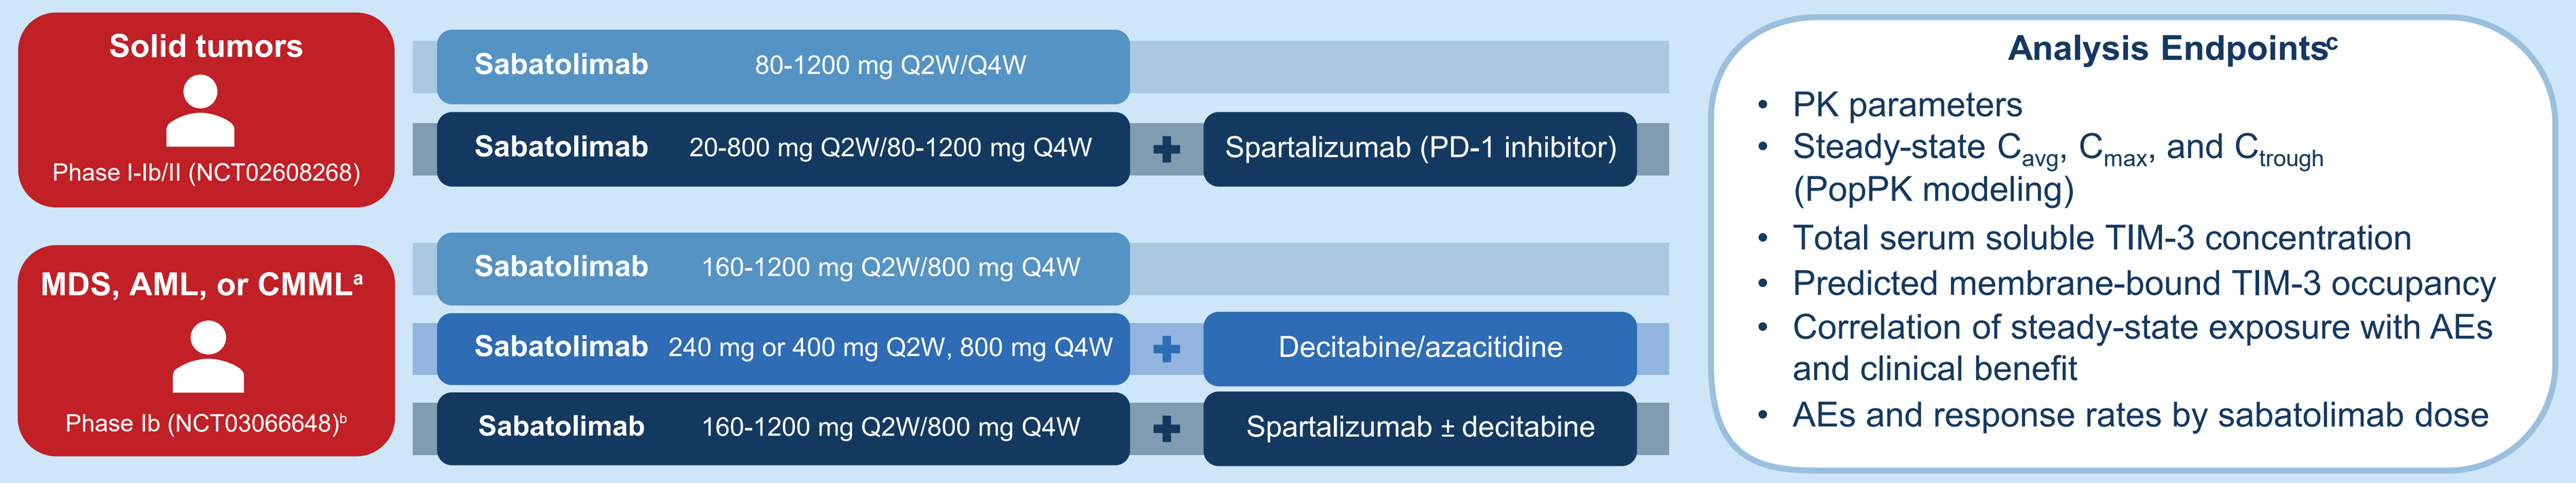
Solid tumors
Phase I-Ib/II (NCT02608268)
Sabatolimab 80-1200 mg Q2W/Q4W
Sabatolimab 20-800 mg Q2W/80-1200 mg Q4W + Spartalizumab (PD-1 inhibitor)
MDS, AML, or CMML[a]
Phase Ib (NCT03066648)[b]
Sabatolimab 160-1200 mg Q2W/800 mg Q4W
Sabatolimab 240 mg or 400 mg Q2W, 800 mg Q4W + Decitabine/azacitidine
Sabatolimab 160-1200 mg Q2W/800 mg Q4W + Spartalizumab ± decitabine
Analysis Endpoints[c]
PK parameters
Steady-state Cavg, Cmax, and Ctrough (PopPK modeling)
Total serum soluble TIM-3 concentration
Predicted membrane-bound TIM-3 occupancy
Correlation of steady-state exposure with AEs and clinical benefit
AEs and response rates by sabatolimab dose

AE, adverse event; AML, acute myeloid leukemia; C_{avg}, average concentration; C_{max}, maximum concentration; C_{trough}, trough concentration; CMML, chronic myelomonocytic leukemia; HR, high-risk; MDS, myelodysplastic syndrome; PD-1, programmed death-1; PK, pharmacokinetics; Pop, population; Q2W, every 2 weeks; Q4W, every 4 weeks; TIM-3, T-cell immunoglobin domain and mucin-3; vHR, very high-risk.
[a] Sabatolimab + decitabine/azacitidine arm included adult patients with vHR/HR-MDS or AML who were ineligible for intensive chemotherapy. [b] Dosing used in vHR/HR-MDS and AML patients was based on findings from the NCT02608268 study. [c] All endpoints had a data cutoff date of November 27, 2019, except for AEs and response rates by sabatolimab dose, which had a data cutoff date of September 22, 2020.

RESULTS

- Saturation of soluble TIM-3 (sTIM-3) was achieved with sabatolimab doses ≥240 mg every 2 weeks (Q2W) and ≥800 mg every 4 weeks (Q4W) (**Figure 4**)
- In patients with solid tumors, sabatolimab doses ≥240 mg Q2W or ≥800 mg Q4W showed:
 - Plateau in accumulated total sTIM-3
 - Proportional dose-exposure relationship
- Similar results were observed in the study in patients with MDS/AML/chronic myelomonocytic leukemia (CMML)

Figure 4. sTIM-3 Levels Over Time by Sabatolimab Dose

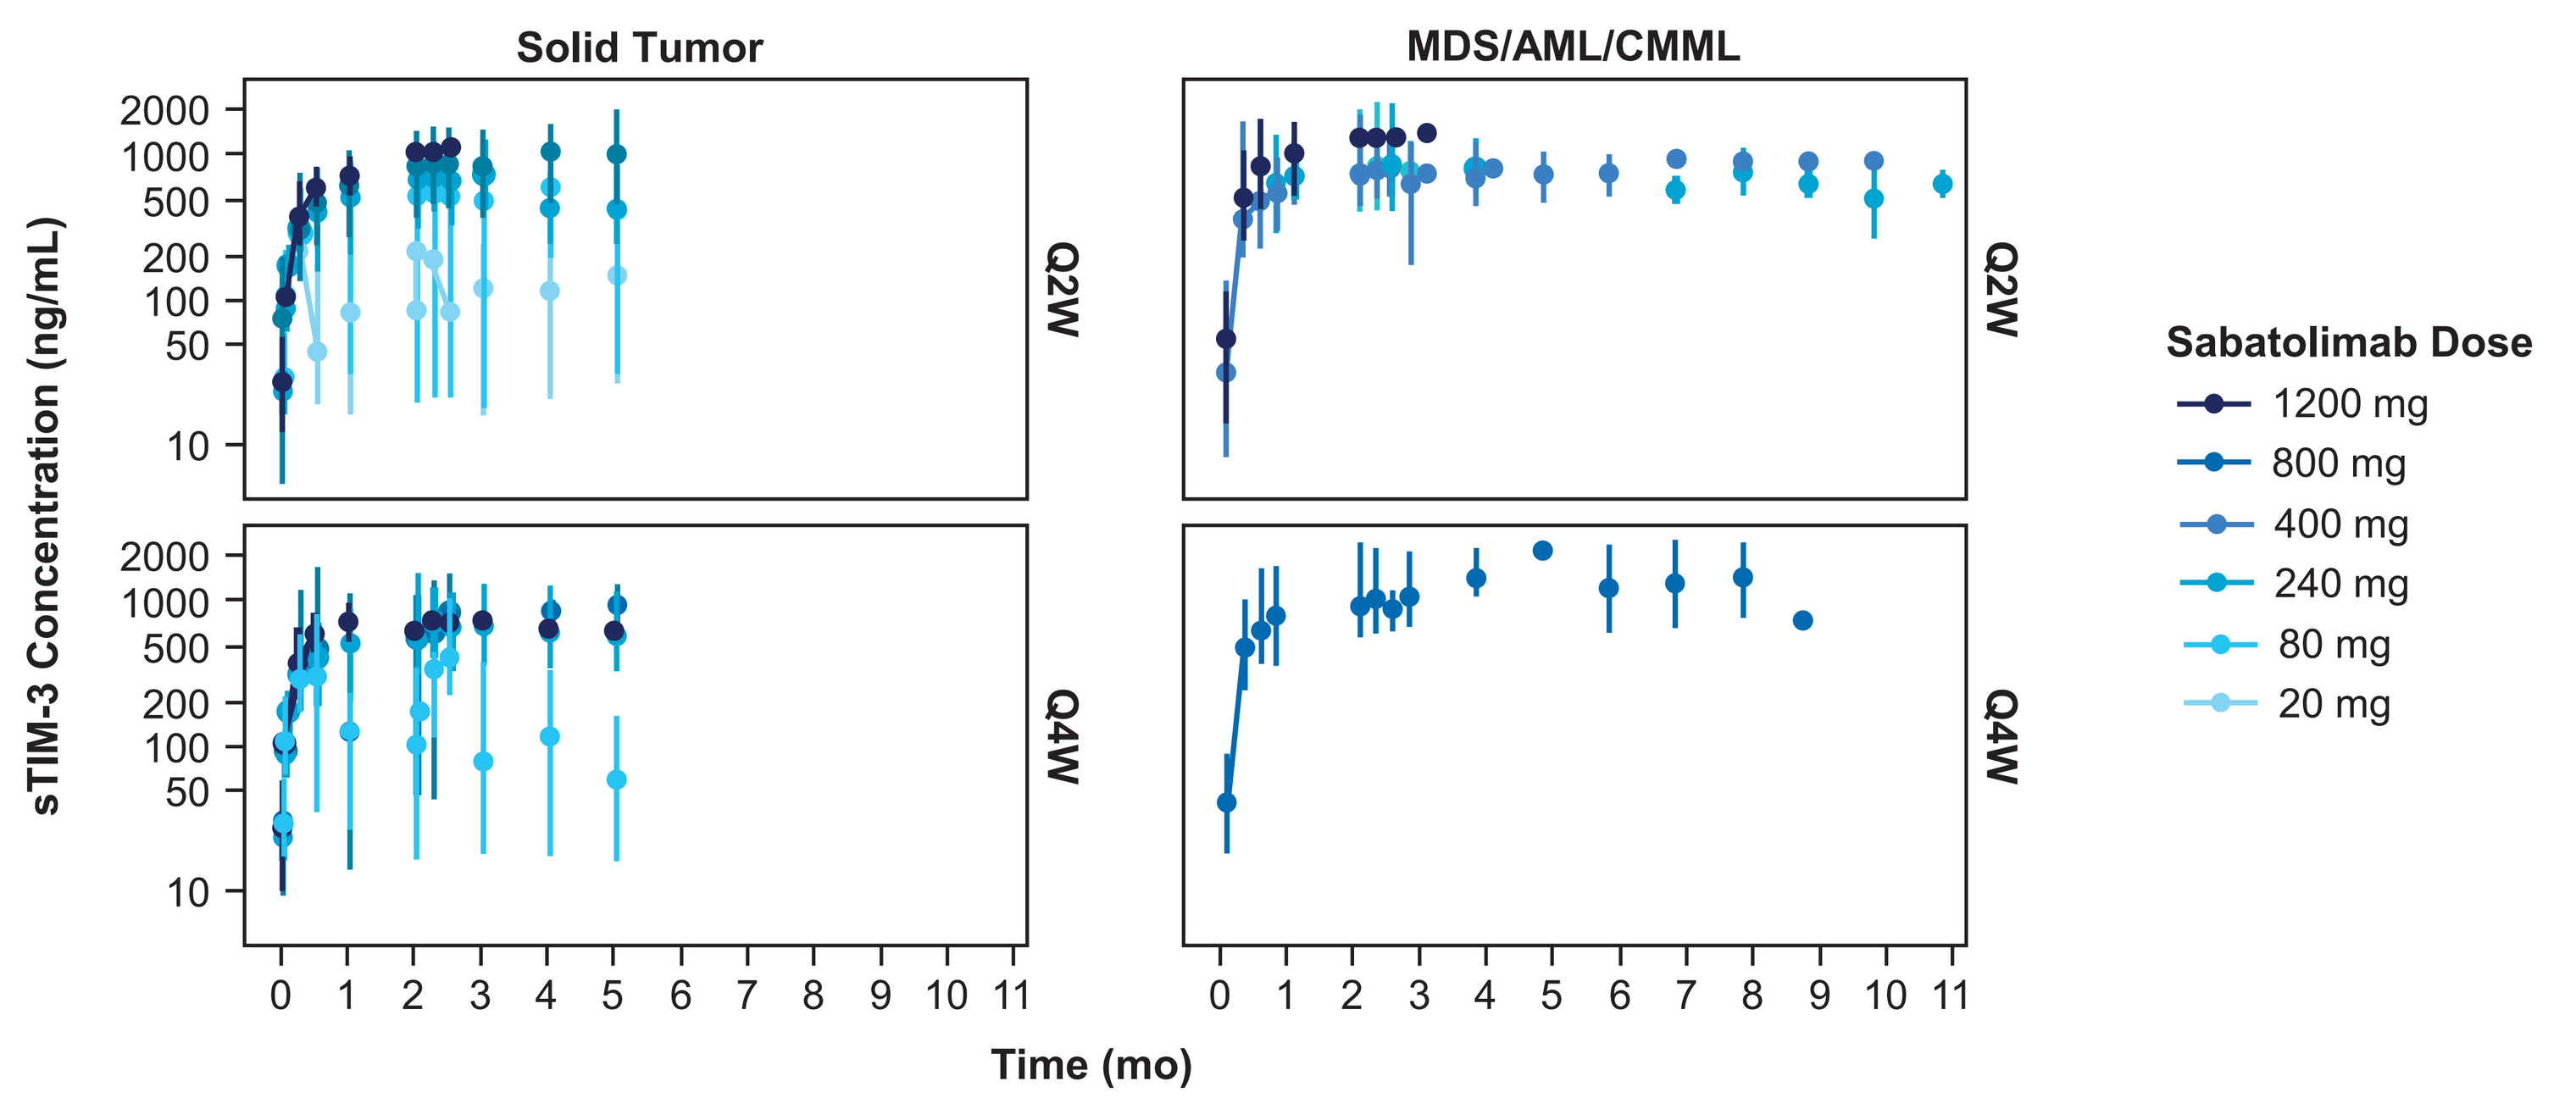
Solid Tumor
MDS/AML/CMML
sTIM-3 Concentration (ng/mL)
2000
1000
500
200
100
50
10
Q2W
Q4W
0 1 2 3 4 5 6 7 8 9 10 11
Time (mo)
Sabatolimab Dose
1200 mg
800 mg
400 mg
240 mg
80 mg
20 mg

AML, acute myeloid leukemia; CMML, chronic myelomonocytic leukemia; MDS, myelodysplastic syndrome; Q2W, every 2 weeks; Q4W, every 4 weeks; sTIM-3, soluble T-cell immunoglobulin domain and mucin domain-3.

Population PK Modeling with Sabatolimab + HMA Regimens

- 400 mg Q2W had the highest trough concentration (C_{trough}) at steady state
- 800 mg was predicted to be an equivalent Q4W dosing regimen
- Both doses had similar steady-state average concentration (C_{avg})
- Receptor occupancy modeling indicated similarly high levels of TIM-3 engagement with sabatolimab 400 mg Q2W and 800 mg Q4W (**Figure 5**)
 - Greater than 95% occupancy rate in bone marrow in ≥95% of patients with very high-risk (vHR)/high-risk (HR)-MDS, AML, or CMML

Figure 5. Simulated Steady-State Occupancy of Membrane-Bound TIM-3 in Bone Marrow[a]

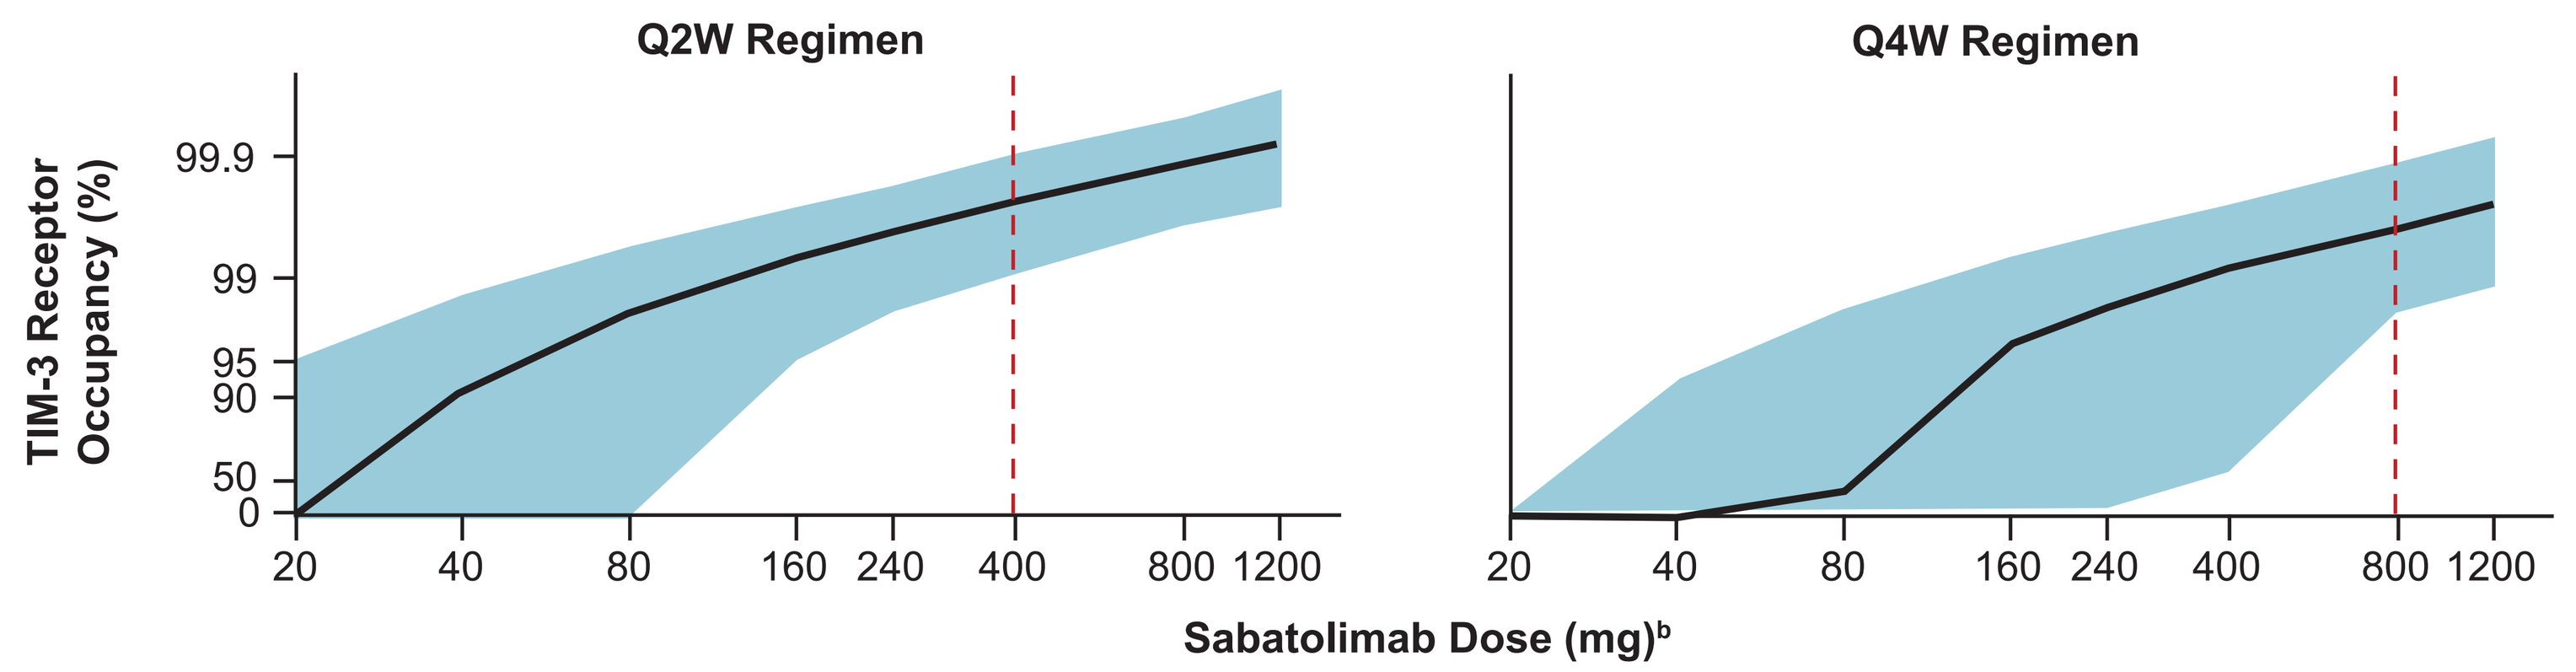
Q2W Regimen
Q4W Regimen
TIM-3 Receptor Occupancy (%)
99.9
99
95
90
50
0
20 40 80 160 240 400 800 1200
Sabatolimab Dose (mg)[b]

TIM-3, T-cell immunoglobin domain and mucin domain-3; Q2W, every 2 weeks; Q4W, every 4 weeks.
[a] Compared to baseline. [b] Shaded region denotes 5% to 95% prediction interval.

- Pharmacokinetic (PK) exposure-response analyses with sabatolimab + hypomethylating agents (HMA) showed similar safety and efficacy across sabatolimab exposure levels (**Figures 6 and 7**)
- Patients were categorized into 4 exposure quartiles based on steady-state maximum concentration (C_{max}) and C_{avg}
 - No relationship between steady-state C_{max} or C_{avg} and treatment-related adverse events (AEs)

Figure 6. PK Exposure-AE Relationship (n=102[a])

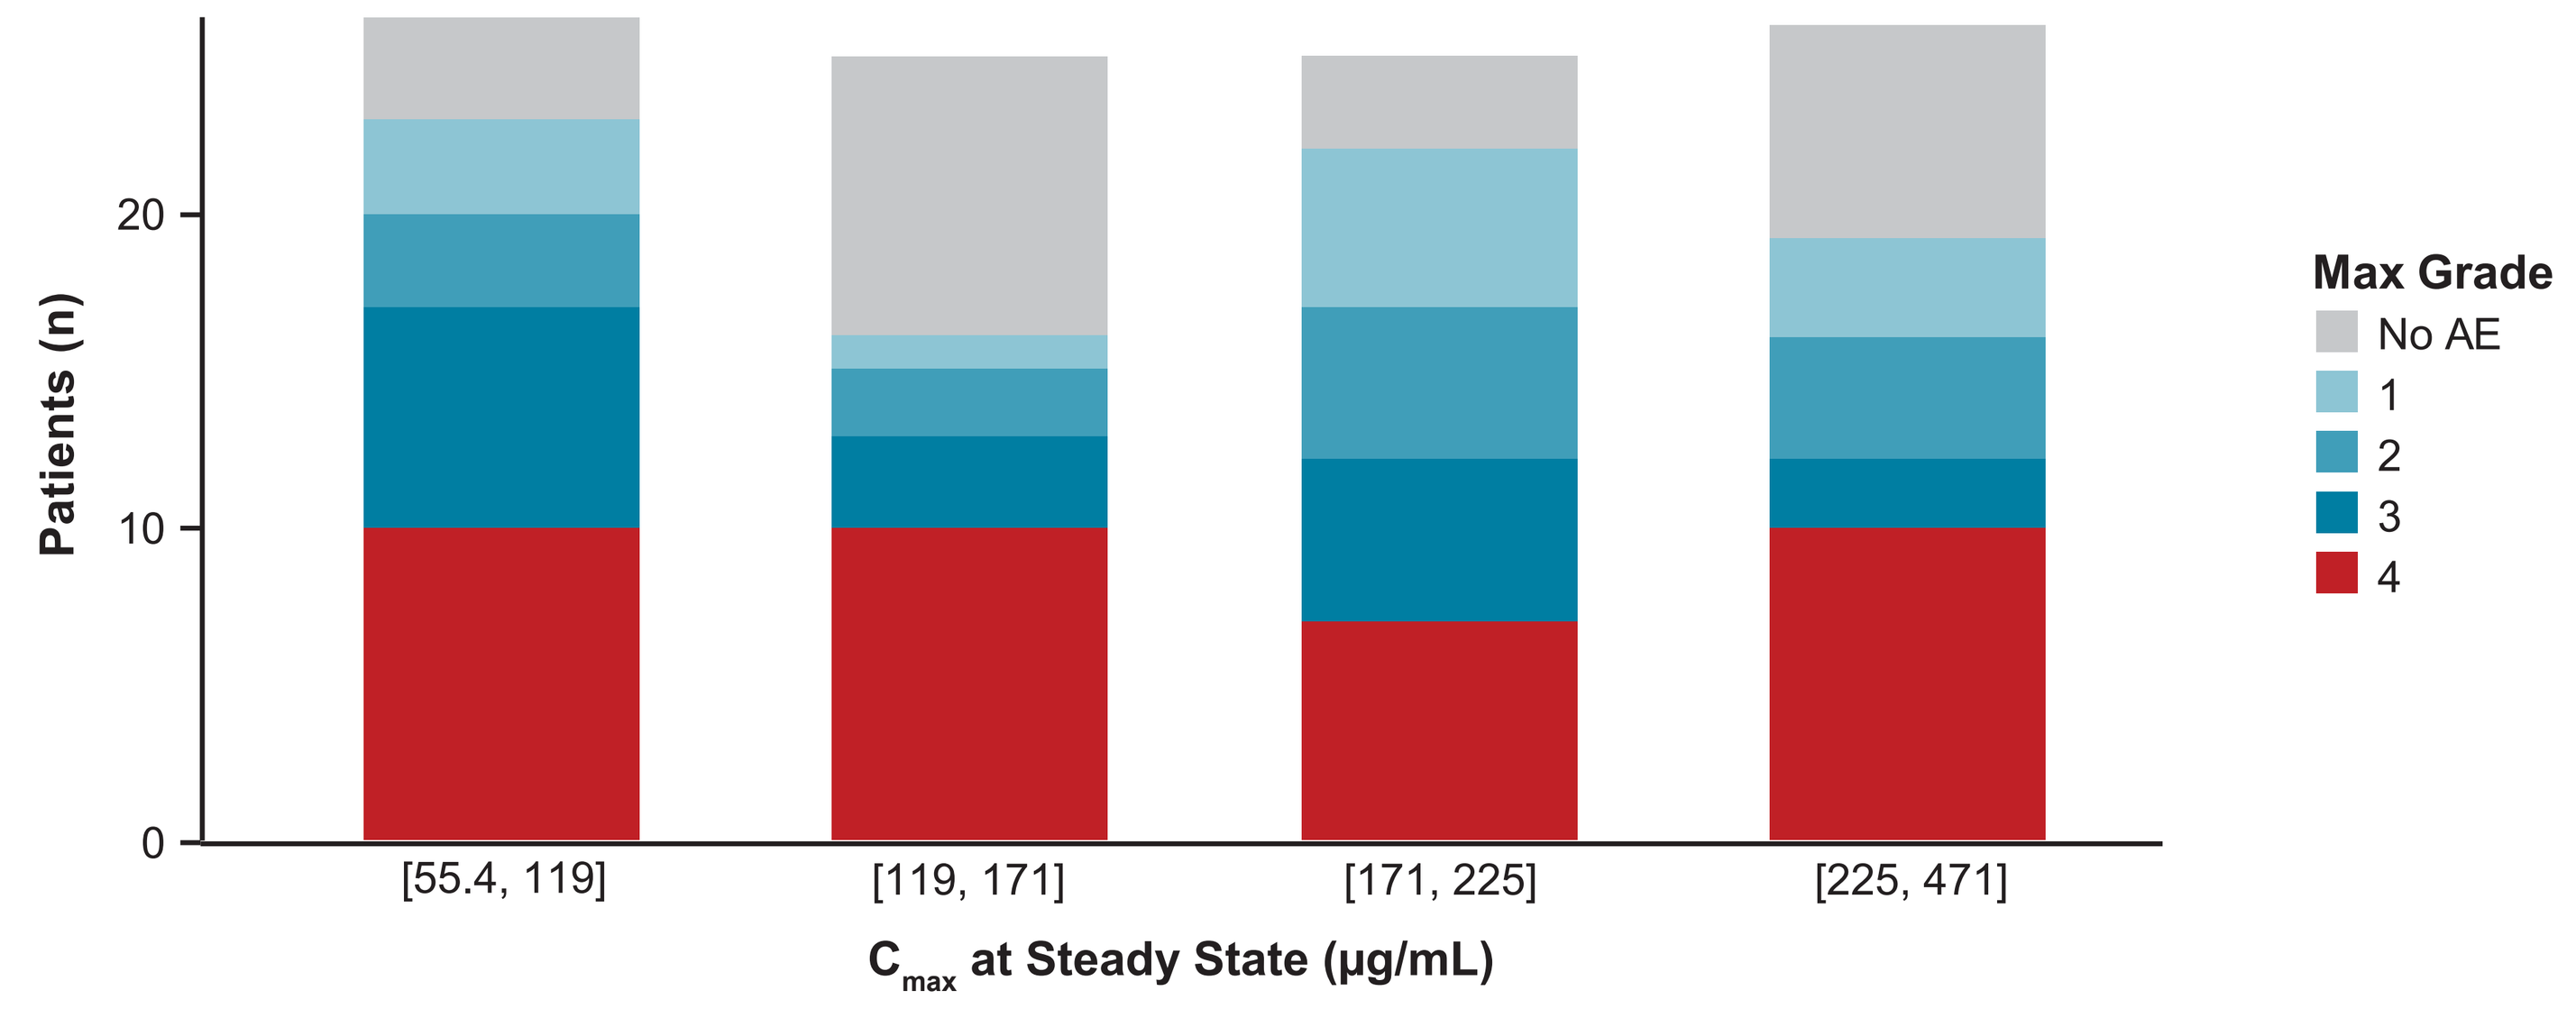
Patients (n)
0
10
20
[55.4, 119]
[119, 171]
[171, 225]
[225, 471]
Cmax at Steady State (μg/mL)
Max Grade
No AE
1
2
3
4

AE, adverse event; AML, acute myeloid leukemia; C_{max}, maximum concentration; CMML, chronic myelomonocytic leukemia; HMA, hypomethylating agent; HR, high-risk; MDS, myelodysplastic syndrome; PK, pharmacokinetics; vHR, very high-risk.
[a] Includes patients with vHR/HR-MDS, CMML, or AML who were treated with sabatolimab + HMA.

- No relationship between steady-state C_{trough} or C_{avg} and % blast cell reduction or clinical benefit (ie, complete remission, marrow complete remission, complete remission with incomplete hematologic recovery, and partial remission) (**Figure 7**)

Figure 7. PK Exposure-Efficacy Relationship (n=92[a])

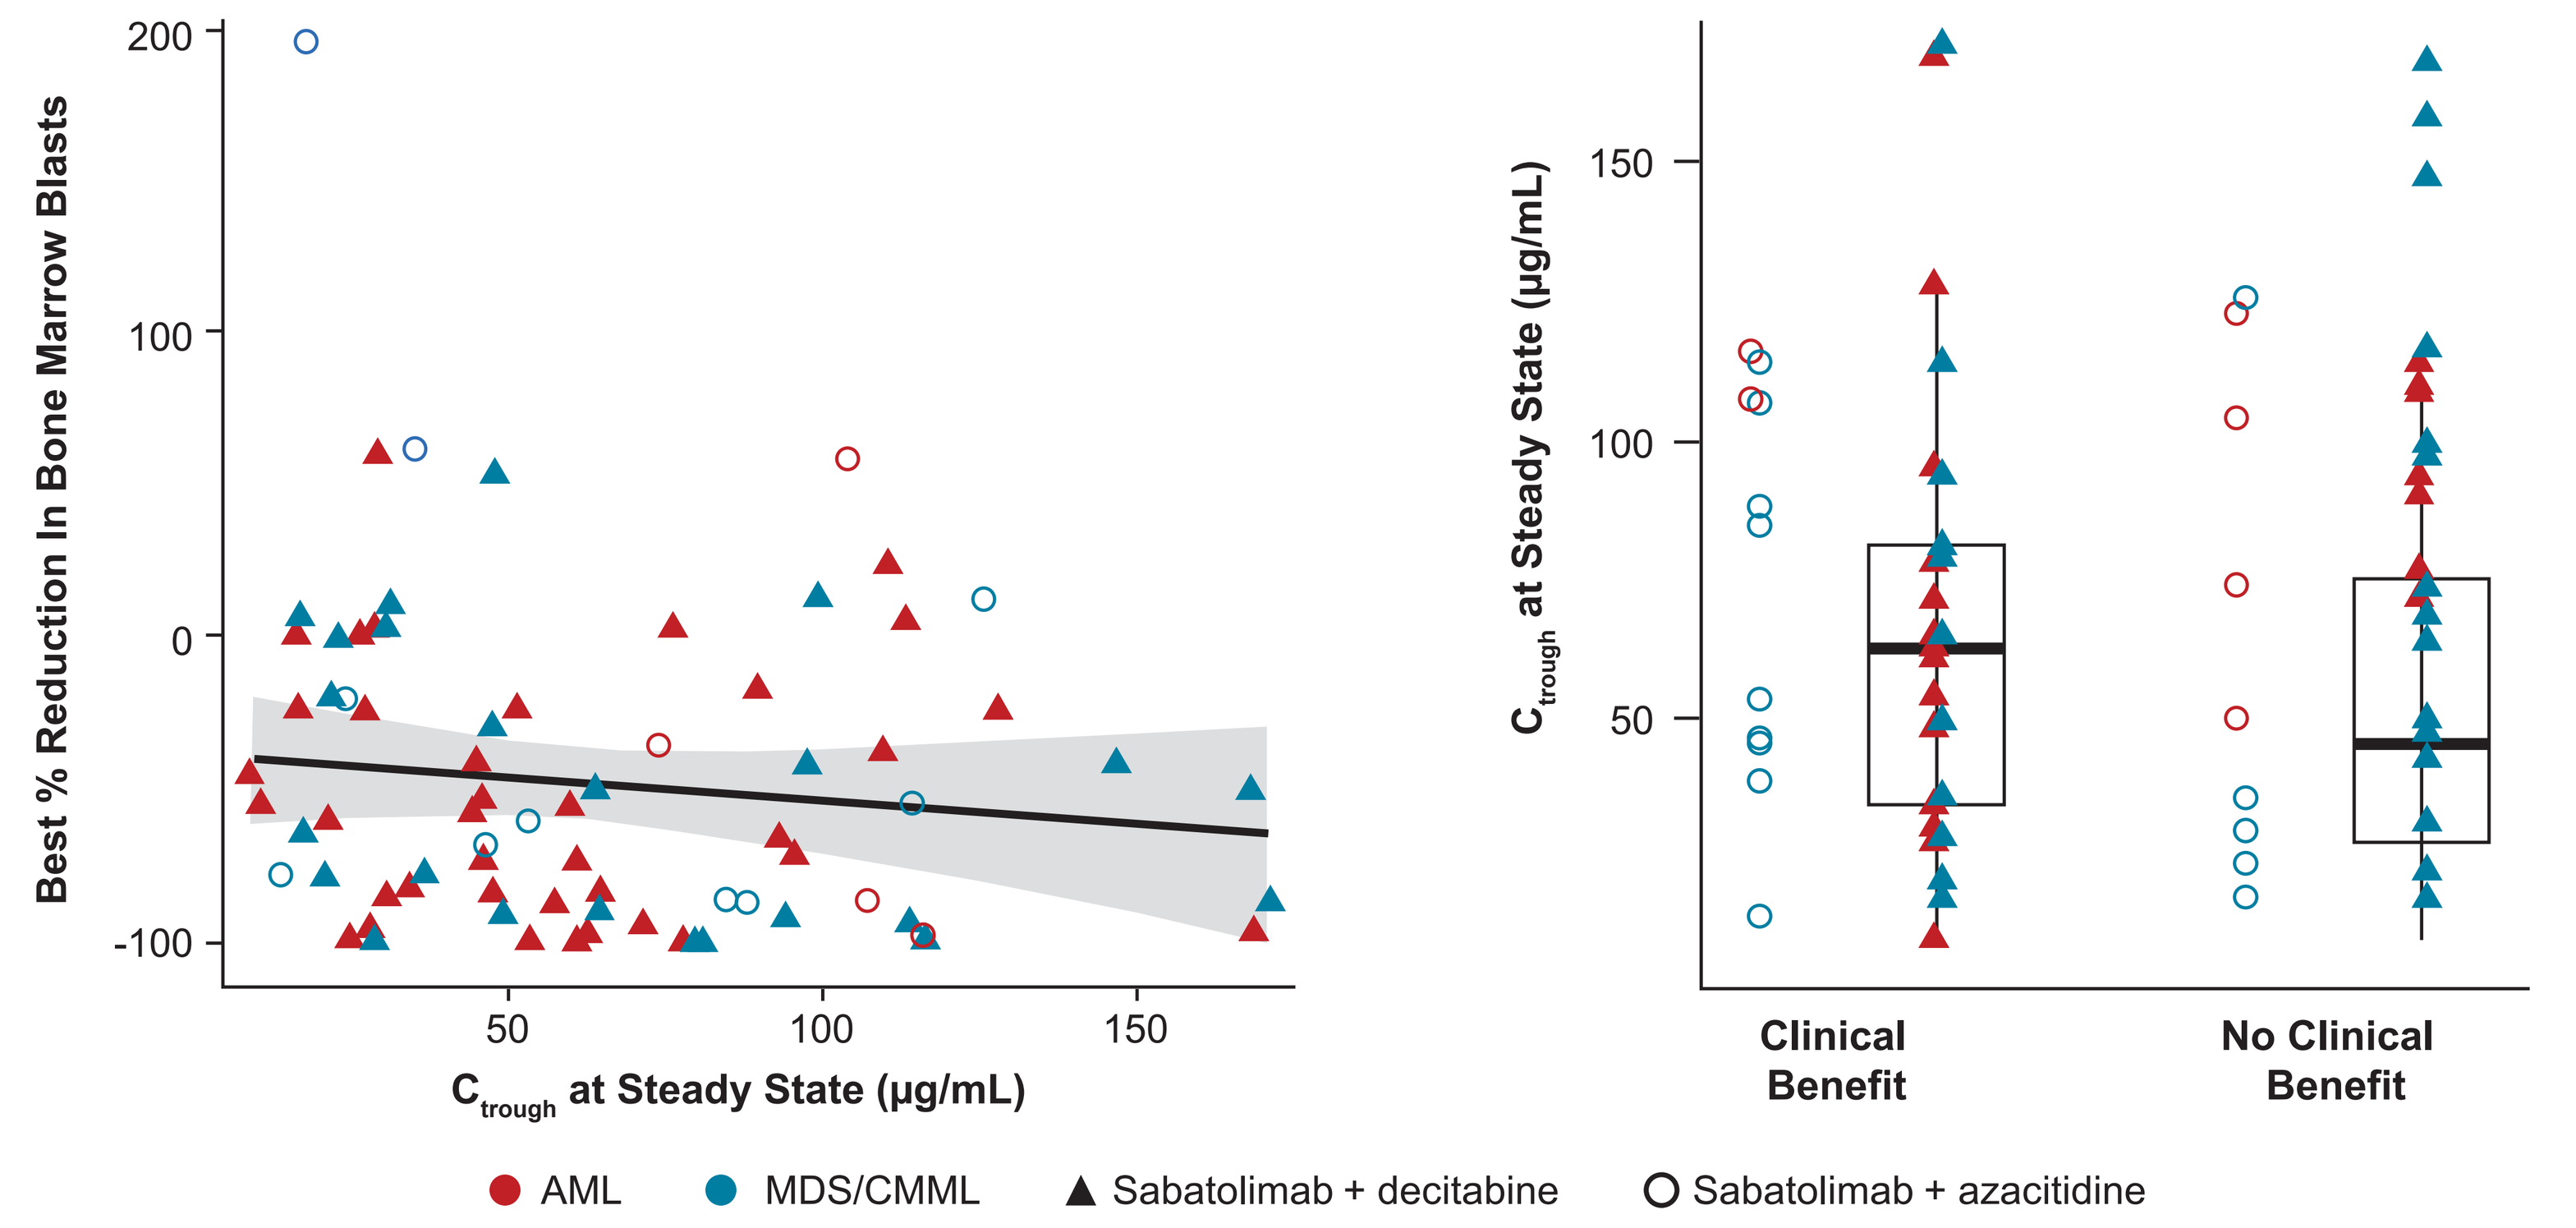
Best % Reduction In Bone Marrow Blasts
200
100
0
-100
50
100
150
Ctrough at Steady State (μg/mL)
Ctrough at Steady State (μg/mL)
150
100
50
Clinical Benefit
No Clinical Benefit
AML
MDS/CMML
Sabatolimab + decitabine
Sabatolimab + azacitidine

AML, acute myeloid leukemia; C_{trough}, trough concentration; CMML, chronic myelomonocytic leukemia; HMA, hypomethylating agent; HR, high-risk; MDS, myelodysplastic syndrome; PK, pharmacokinetics; vHR, very high-risk.
[a] Includes patients with vHR/HR-MDS, CMML, or AML who were treated with sabatolimab + HMA.

- Rates of clinically significant (grade ≥3) treatment-emergent AEs (TEAEs) and possible immune-mediated AEs (imAEs) related to treatment did not appear to be dose dependent (**Table 1**)

Table 1. Clinical Safety by Sabatolimab Dose (n=118)

	vHR/HR-MDS and AML		
Sabatolimab dose	240 mg Q2W	400 mg Q2W	800 mg Q4W
Safety population, n	30	50	38
Most common[a] TEAEs grade ≥3, n (%)			
Neutropenia	15 (50.0)	20 (40.0)	17 (44.7)
Thrombocytopenia	13 (43.3)	16 (32.0)	20 (52.6)
Febrile neutropenia	17 (56.7)	14 (28.0)	14 (36.8)
Anemia	9 (30.0)	12 (24.0)	14 (36.8)
Pneumonia	2 (6.7)	8 (16.0)	5 (13.2)
Treatment-related possible imAEs grade ≥3,[b] n (%)	4 (13.3)	0	2 (5.3)

AML, acute myeloid leukemia; HR, high-risk; imAE, immune-mediated adverse event; MDS, myelodysplastic syndrome; Q2W, every 2 weeks; Q4W, every 4 weeks; TEAE, treatment-emergent adverse event; vHR, very high-risk.
[a] Reported in ≥10% of patients in either the vHR/HR-MDS or AML cohort. [b] 8 events occurred in 6 patients.

- To date, among the 3 sabatolimab dosing regimens tested (**Figure 8**):
 - No notable differences in efficacy
 - No significant association with duration of response (Cox model with dose as a covariate)
- Sabatolimab + HMA was safe overall, with encouraging efficacy across doses in vHR/HR-MDS or AML

Figure 8. Clinical Efficacy by Sabatolimab Dose (n=104)

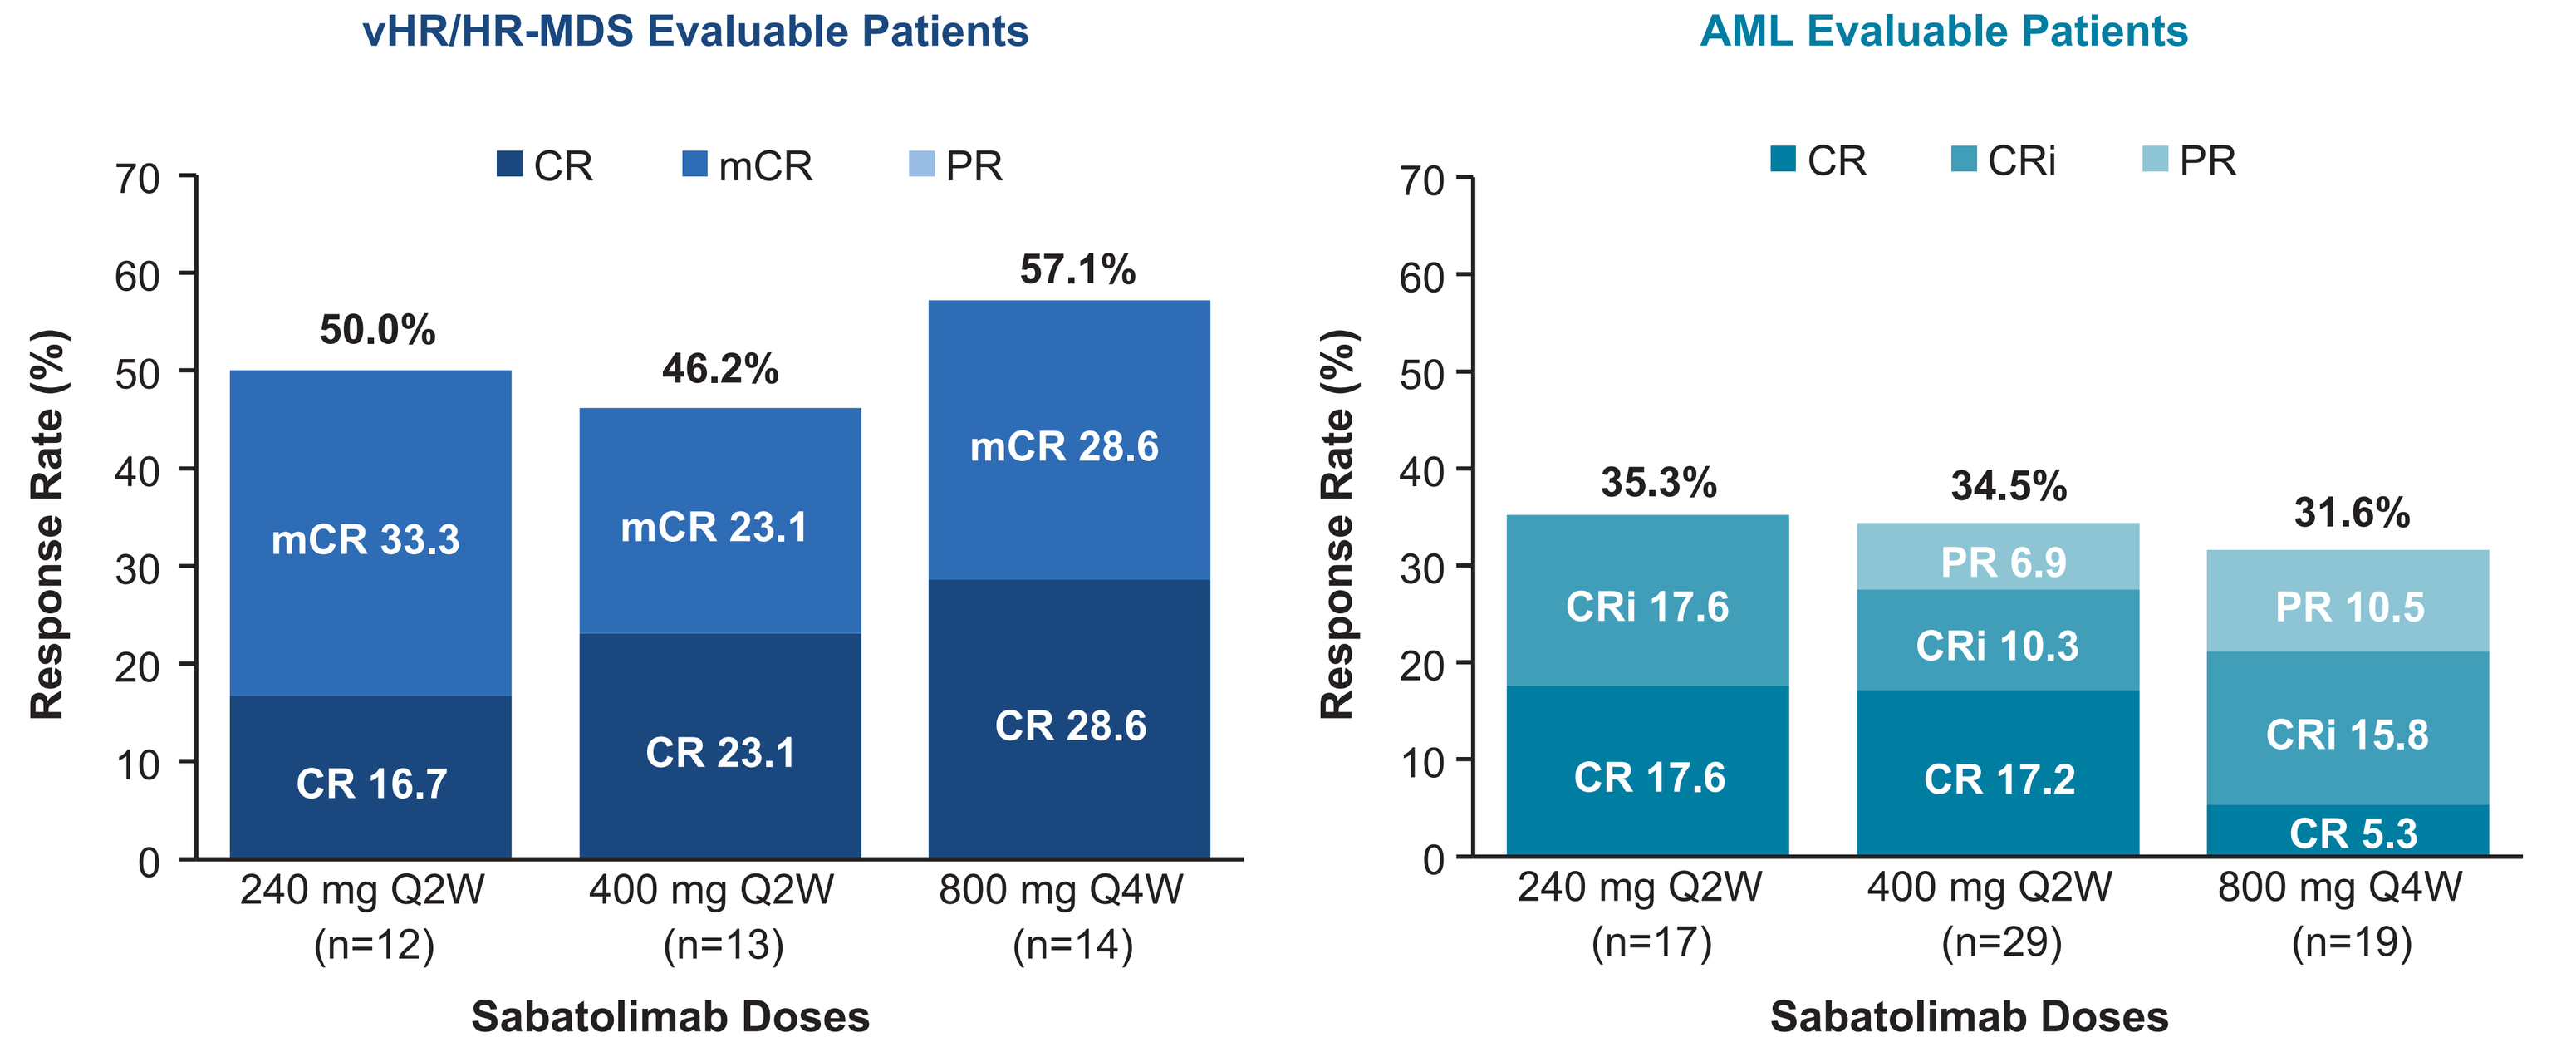
vHR/HR-MDS Evaluable Patients
AML Evaluable Patients
CR
mCR
PR
CRi
Response Rate (%)
0 10 20 30 40 50 60 70
50.0%
mCR 33.3
CR 16.7
46.2%
mCR 23.1
CR 23.1
57.1%
mCR 28.6
CR 28.6
240 mg Q2W (n=12)
400 mg Q2W (n=13)
800 mg Q4W (n=14)
35.3%
CRi 17.6
CR 17.6
34.5%
PR 6.9
CRi 10.3
CR 17.2
31.6%
PR 10.5
CRi 15.8
CR 5.3
240 mg Q2W (n=17)
400 mg Q2W (n=29)
800 mg Q4W (n=19)
Sabatolimab Doses

AML, acute myeloid leukemia; CR, complete remission; CRi, complete remission with incomplete hematologic recovery; HR, high-risk; mCR, marrow complete remission; MDS, myelodysplastic syndrome; PR, partial remission; Q2W, every 2 weeks; Q4W, every 4 weeks; vHR, very high-risk.

Acknowledgements

The authors thank the patients who participated in the trial and their families.

Financial support for medical editorial assistance was provided by Novartis Pharmaceuticals Corporation. The authors thank Kendall Foote, PhD, of Medical Expressions for medical editorial assistance with this poster presentation.

Disclosures

GG-M reports consultancy for Acceleron Pharmaceuticals, Astex Pharmaceuticals, Bristol-Myers Squibb, Celgene, Helsinn Therapeutics, Jazz Pharmaceuticals, and Genentech; honoraria for AbbVie, Acceleron Pharmaceuticals, Astex Pharmaceuticals, Celgene, and Helsinn Therapeutics; research funding from AbbVie, Amphivena Therapeutics, Astex Pharmaceuticals, Bristol-Myers Squibb, Celgene, Helsinn Therapeutics, H3 Biomedicine, Merck, Novartis, Genentech, and Onconova; membership on an entity's Board of Directors or advisory committees for Genentech. **AHW** reports consultancy for Servier, BMS; honoraria for AbbVie, Servier, Novartis, AstraZeneca, Pfizer, BMS, Janssen, Amgen, Astellas, Genentech, and Celgene; research funding from Abbvie, Servier, Novartis, AstraZeneca, BMS, Amgen, and Celgene; speakers bureau for AbbVie, BMS, and Novartis; patents and royalties from Walter and Eliza Hall Institute of Medical Research; membership on an entity's Board of Directors or advisory committees for Astellas, Genentech, and Celgene. **KP** reports consultancy for Novartis; honoraria and research funding from Novartis and BMS/Celgene. **SK** reports honoraria, membership on an entity's Board of Directors or advisory committee and research funding for Novartis. **MW** reports honoraria for MacroGenics. **JJ** reports honoraria for Novartis, Pfizer, and Incyte; research funding from Novartis, and BMS, membership on an entity's Board of Directors or advisory committees for Novartis, Pfizer, AbbVie, and Incyte; Founder of the HematologyApp which is supported by Janssen, BMS Incyte, MDS, Pfizer, Daiichi-Sankyo, Roche and Takeda. **ET** reports consultancy for AbbVie, and Notable Labs; research funding for Incyte; membership on an entity's Board of Directors or advisory committees for Agios, Genentech, AbbVie, Astellas, and Daiichi Sankyo; current equity holder in private company with Notable Labs. **RN** reports current spouse employment with Sanofi-Genzyme; prior spouse employment within 24 months with Takeda; prior spouse employment within 24 months and prior spouse equity divested within past 24 months with Genentech. **MK** reports consultancy with Astellas; research funding with AbbVie; membership on an entity's Board of Directors or advisory committees with Jazz Pharma, Astellas, Novartis, and Celgene; speakers bureau with Novartis, and Celgene. **OO** reports honoraria for Novartis, Amgen, Celgene, Fusion Pharma, and Incyte; research funding from Amgen, Celgene, and Incyte. **MLR** reports employment at Novartis and consultancy for Qiagen. **AMB** reports consultancy for Acceleron Pharma Inc., Biogen, Celgene/BMS, Forty Seven, Inc., Jazz Pharma, Novartis, Takeda, and Xcenda, research funding from Celgene/BMS, Novartis, Takeda, GSK, Janssen, and AstraZeneca. **UB** reports research funding from Takeda, Novartis, Jazz Pharmaceuticals, and Pfizer; membership on an entity's Board of Directors or advisory committees for Genentech, Daiichi Sankyo, Takeda, Novartis, and Pfizer; investigator in AbbVie-funded clinical trials. **SX, AL, AS, PN, NZ, AM,** and **HS** are employees at Novartis. All other authors had nothing to disclose.

References

1. Wolf Y, et al. *Nat Rev Immunol*. 2020;20(3):173-185. **2.** Acharya N, et al. *J Immunother Cancer*. 2020;8(1):e000911. **3.** Haubner S, et al. *Leukemia*. 2019;33(1):64-74. **4.** Asayama T, et al. *Oncotarget*. 2017;8(51):88904-88917. **5.** Kikushige Y, et al. *Cell Stem Cell*. 2015;17(3):341-352. **6.** Mach N, et al. *Ann Oncol*. 2019;30(suppl 5):abstract 1202P. **7.** Borate U, et al. *HemaSphere*. 2020;4(suppl 1):abstract S185. 8. Borate U, et al. *EHA 2020*. Oral presentation.
9. Sabatos-Peyton C, et al. *SITC 2020*. Abstract 439.