MDS-364

Amer M. Zeidan, amer.zeidan@yale.edu

STIMULUS MDS-US Trial in Progress: Evaluating Sabatolimab in Combination With Hypomethylating Agents in Patients With Intermediate-, High-, or Very High-Risk Myelodysplastic Syndromes

Amer M. Zeidan[1], Amy DeZern[2], Uma Borate[3], Krissy Kobata[4], Islam Sadek[5], J. Randy Sabo[5], Das Purkayastha[5], Pedro Marques Ramos[6], Haiying Sun[5], Roger Lyons[7], Guillermo Garcia-Manero[8]

[1]Yale University, New Haven, CT, USA; [2]Johns Hopkins University, Baltimore, MD, USA; [3]Oregon Health & Science University, Portland, OR, USA; [4]Los Angeles, CA, USA; [5]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [6]Novartis Pharma AG, Basel, Switzerland; [7]Texas Oncology and US Oncology Research, San Antonio, TX, USA; [8]The University of Texas MD Anderson Cancer Center, Houston, TX, USA

Scan to obtain:
- Poster
- Supplementary Podcast

http://novartis.medicalcongressposters.com/Default.aspx?doc=6f893

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

CONCLUSIONS

- Recruitment is underway in the Phase II STIMULUS MDS-US trial evaluating the safety and efficacy of sabatolimab in combination with US FDA–approved HMAs in patients with intermediate-, high-, or very high-risk MDS
- Approximately 90 patients will be enrolled across multiple sites in the United States
- Sabatolimab is an investigational immuno-myeloid therapy that targets TIM-3 on immune and leukemic cells; sabatolimab has demonstrated an encouraging safety and tolerability profile and emerging durable responses in patients with higher-risk MDS

This study is sponsored by Novartis Pharmaceuticals Corporation.
Presented at the Society of Hematologic Oncology Ninth Hybrid Annual Meeting; September 8-11, 2021.

INTRODUCTION

Myelodysplastic Syndromes

- Myelodysplastic syndromes (MDS) are a heterogenous group of hematopoietic diseases characterized by a dysfunctional immune system and leukemic stem cell (LSC) proliferation; they are associated with impaired bone marrow function, persistent cytopenia, and a risk of progression to acute myeloid leukemia (AML)[1]
- Hypomethylating agents (HMAs) are approved for the frontline management of patients with MDS classified as intermediate-, high-, or very high-risk by the revised International Prognostic Scoring System (IPSS-R) and who do not qualify for hematopoietic stem cell transplant or intensive chemotherapy[2]
 - Fifty percent of HMA-treated patients experience primary failure, and most responders have disease progression within 1 to 2 years[1]
- Novel therapies that provide improved durable outcomes with favorable safety profiles are needed for these patients

TIM-3 Biology in MDS

- T-cell immunoglobulin domain and mucin domain-3 (TIM-3) is a novel immune target with a unique role in myeloid malignancies and is a key regulator of innate and adaptive immune responses[3,4] (**Figure 1**)
- TIM-3 is aberrantly expressed on LSCs and blasts, but not on normal hematopoietic stem cells,[3-7] making it a potential target in MDS/AML[4,6,8]
- TIM-3/galectin-9 interaction forms an autocrine stimulatory loop promoting LSC self-renewal[9,10]

Figure 1. TIM-3 is an Inhibitory Receptor Expressed on Immune and Leukemic Cells

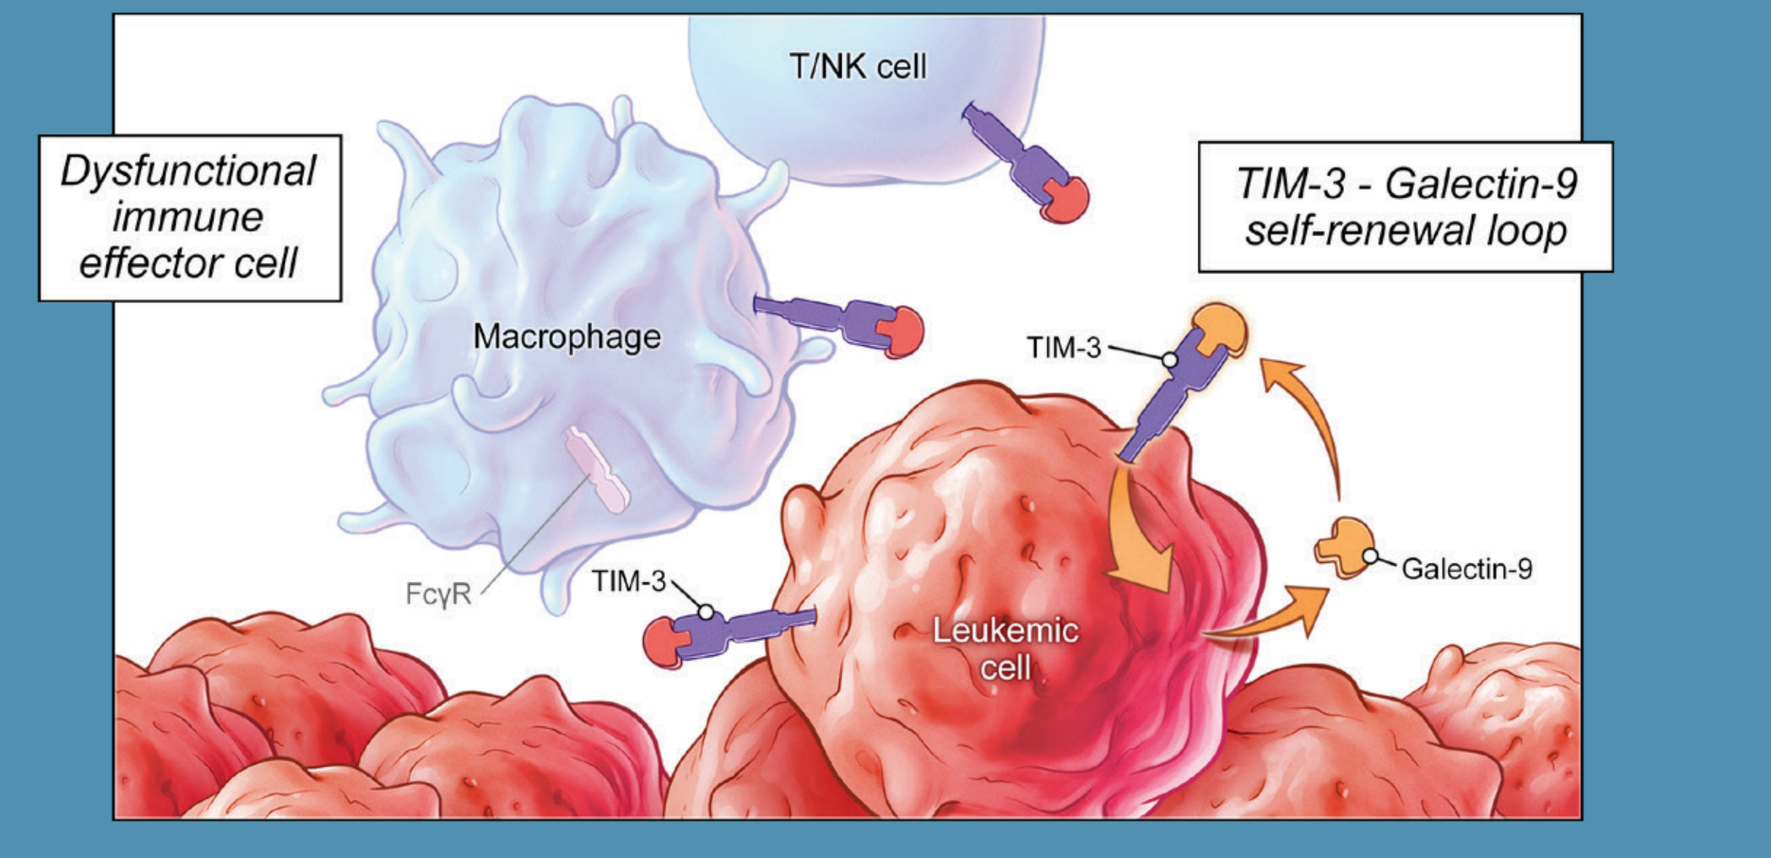
T/NK cell
Dysfunctional immune effector cell
TIM-3 - Galectin-9 self-renewal loop
Macrophage
TIM-3
FcγR
TIM-3
Galectin-9
Leukemic cell

FcγR, Fc gamma receptor; NK, natural killer; TIM-3, T-cell immunoglobulin domain and mucin domain-3.

SABATOLIMAB OVERVIEW

- Sabatolimab is an investigational immuno-myeloid therapy that targets TIM-3 on immune and leukemic cells (**Figure 2**)
- Sabatolimab has been shown to:
 - Bind TIM-3 on immune cells, enhancing antileukemic immune activation and phagocytic killing of LSCs and blasts[11-13]
 - Directly target TIM-3 on LSCs, inhibiting TIM-3/galectin-9–driven self-renewal[11,12]
- In a Phase Ib study (NCT03066648), sabatolimab + HMAs demonstrated an encouraging safety and tolerability profile and emerging durable responses in patients with higher-risk MDS[14]
 - Overall response rate: 58%
 - Median duration of response: 16.1 months
- The STIMULUS immuno-myeloid clinical trial program is investigating the safety and efficacy of sabatolimab in myeloid malignancies, including high-risk MDS and AML[15]
 - The STIMULUS MDS-US trial has been designed to assess the safety and efficacy of sabatolimab in combination with United States Food and Drug Administration (US FDA)–approved HMAs of investigator's choice

Figure 2. Sabatolimab Targets TIM-3 on Immune and Leukemic Cells: A Novel Immuno-Myeloid Therapy

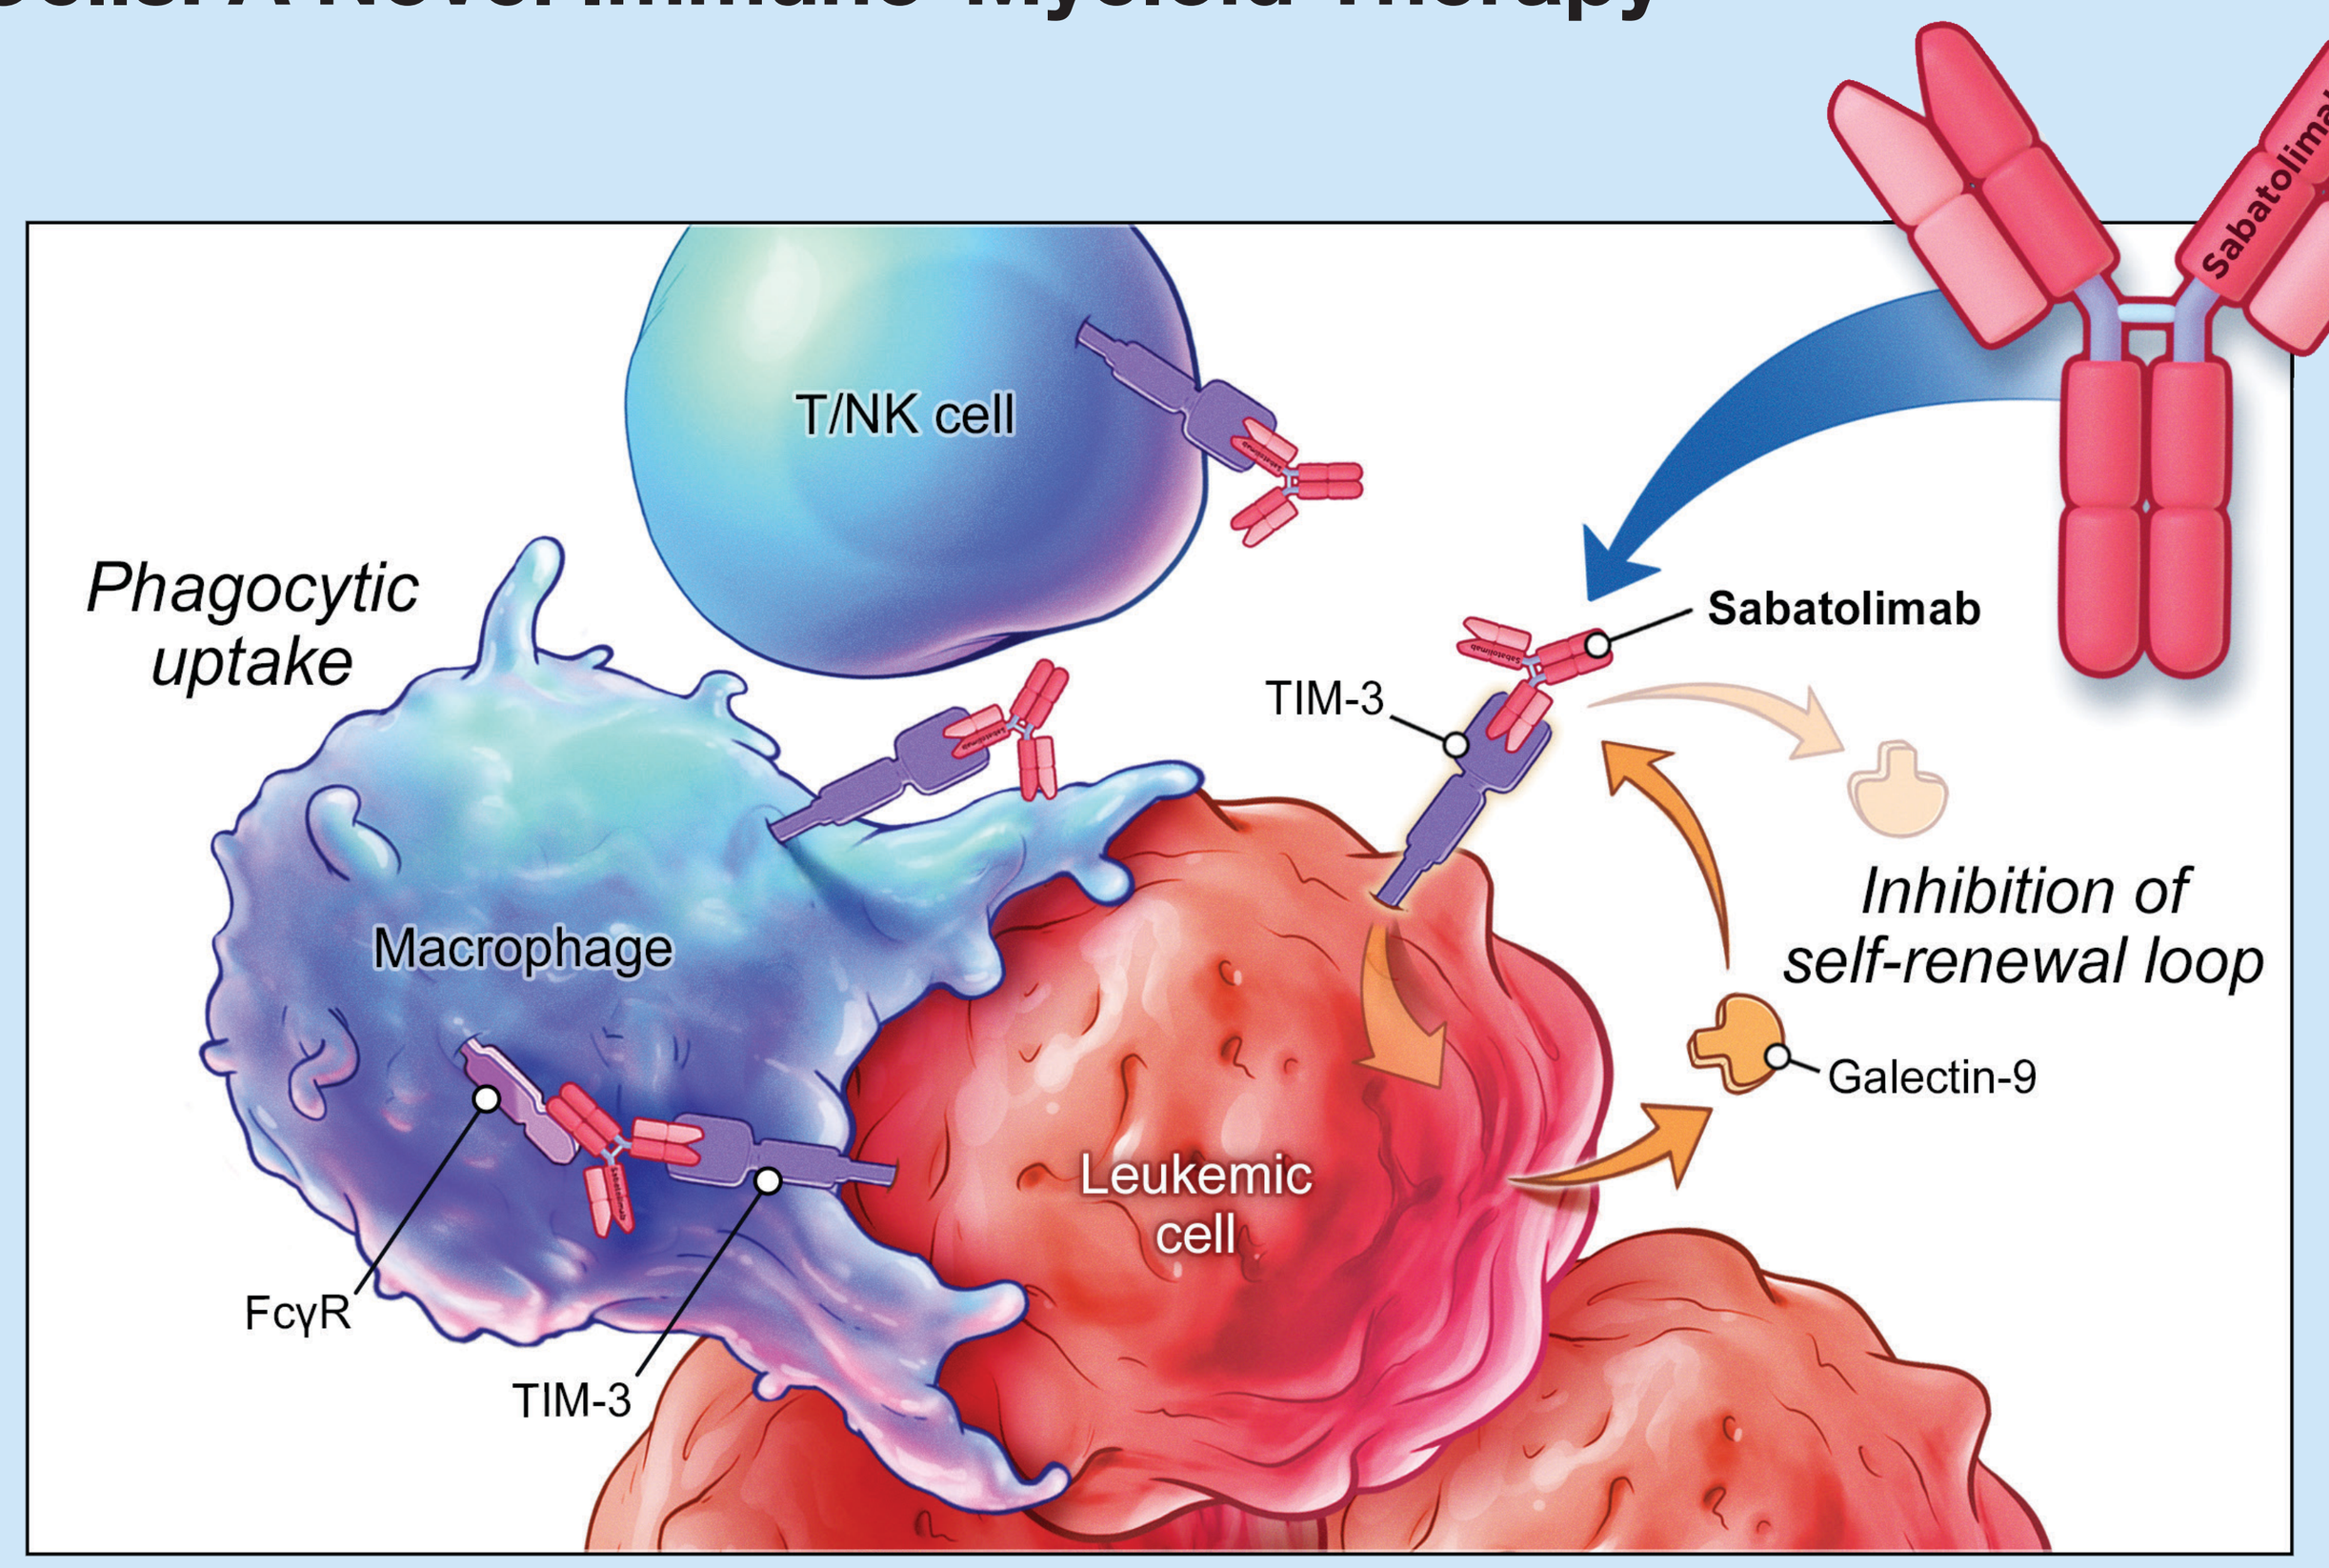
Sabatolimab
T/NK cell
Phagocytic uptake
Sabatolimab
TIM-3
Inhibition of self-renewal loop
Macrophage
Galectin-9
Leukemic cell
FcγR
TIM-3

FcγR, Fc gamma receptor; NK, natural killer; TIM-3, T-cell immunoglobulin domain and mucin domain-3.

METHODS

- STIMULUS MDS-US (NCT04878432) is a Phase II, nonrandomized, single-arm, open-label, multicenter study (**Figure 3**)
 - Patients will receive sabatolimab in combination with investigator's choice of HMA
 - Sabatolimab 800 mg will be administered by infusion over 30 minutes every 4 weeks
 - For intravenous and subcutaneous HMAs, sabatolimab can be administered on any day between days 5 and 8
- The HMA dosing schedule is explained in **Table 1**
- Study endpoints are shown in **Table 2**
- Key eligibility criteria are described in **Table 3**

Figure 3. Study Design

Screening	Core Phase: 28-day cycles for 24 months	Extension Phase: 12 months	
Patients aged ≥18 years with morphologically-confirmed MDS[a] *(N = 90)*	Sabatolimab 800 mg IV Q4W[b] + HMA of investigator's choice (IV, SC, or oral)	EOT or continued treatment per investigator decision	**Safety Follow-Up: up to 150 days**

EOT, end of treatment; HMA, hypomethylating agent; IPSS-R, revised International Prognostic Scoring System; IV, intravenous; MDS, myelodysplastic syndrome; Q4W, once every 4 weeks; SC, subcutaneous.
[a] IPSS-R prognostic risk score intermediate, high, or very high. [b] 30-min infusion.

Table 1. HMA Dose Schedule

HMA	Oral decitabine and cedazuridine	IV decitabine	IV or SC azacitidine
Dose	100 mg/35 mg days 1-5	20 mg/m² days 1-5	75 mg/m² days 1-7 or days 1-5 and 8+9

HMA, hypomethylating agent; IV, intravenous; SC, subcutaneous.

Table 2. Study Endpoints

Primary Endpoint
• Number of treatment-emergent AEs and serious AEs
Secondary Endpoints
• Complete remission rate according to International Working Group for MDS (2006) as per investigator assessment by 12 months
• Progression-free survival
• Overall survival
• Leukemia-free survival
• Percentage of patients with complete response, marrow complete response, and/or partial response
• Duration of complete remission
• Time to remission
• Percentage of patients with improvement in red blood cell/platelet transfusion independence

AE, adverse event; MDS, myelodysplastic syndrome.

Table 3. Key Eligibility Criteria

Inclusion Criteria
• Age ≥18 years
• Morphologically-confirmed diagnosis of MDS with 1 of the following prognostic risk categories, based on the IPSS-R[16]: very high (>6 points), high (>4.5-6 points), or intermediate (>3-4.5 points)
• Not suitable for immediate myeloablative chemotherapy or HSCT
• ECOG performance status of 0, 1, or 2
• AST and ALT ≤3 × ULN
• Total bilirubin ≤2 × ULN
• eGFR ≥30 mL/min/1.73 m²

ALT, alanine aminotransferase; AST, aspartate aminotransferase; ECOG, Eastern Cooperative Oncology Group; eGFR, estimated glomerular filtration rate; HSCT, hematopoietic stem cell transplant; IPSS-R, revised International Prognostic Scoring System; MDS, myelodysplastic syndrome; ULN, upper limit of normal.

Table 3. Key Eligibility Criteria, Cont'd

Exclusion Criteria
• Prior exposure to TIM-3–directed therapy at any time; prior therapy with immune checkpoint inhibitors or cancer vaccines is allowed only if the last dose was administered >4 months prior to enrollment
• Previous treatment of intermediate-, high- or very high-risk MDS with chemotherapy or other antineoplastic agents including lenalidomide and HMAs such as decitabine, azacytidine, or oral decitabine and cedazuridine
– Patients who have received up to 1 cycle of HMAs can be included and previous hydroxyurea treatment is permitted
• Previous diagnoses based on 2016 WHO classifications[16]:
– AML, including APL and extra-medullary AML
– CMML or primary or secondary MF
• History of organ transplant or allogenic HSCT
• Prior malignancies, except:
– Lower-risk MDS treated by supportive care (eg, growth factors, TGFβ agents) or untreated
– Lower-risk MDS treated adequately with, then failed lenalidomide
– Adequately treated malignancies for which no anticancer systemic therapy is ongoing or required during the study; patients receiving adjuvant therapy, such as hormone therapy, are eligible
• Patients with MDS with IPSS-R ≤3[16]

AML, acute myeloid leukemia; APL, acute promyelocytic leukemia; CMML, chronic myelomonocytic leukemia; HSCT, hematopoietic stem cell transplant; HMA, hypomethylating agent; IPSS-R, revised International Prognostic Scoring System; MDS, myelodysplastic syndrome; MF, myelofibrosis; TGFβ, transforming growth factor beta; TIM-3, T-cell immunoglobulin domain and mucin domain-3; WHO, World Health Organization.

Acknowledgements

The authors thank all study participants and their families, the study investigators, and staff participating at study sites.

Financial support for medical editorial assistance was provided by Novartis Pharmaceuticals Corporation. The authors thank Amanda Martin, PhD, from Medical Expressions for medical editorial assistance with this poster presentation.

Disclosures

AMZ received research funding (institutional) from Celgene/BMS, Abbvie, Astex, Pfizer, Medimmune/AstraZeneca, Boehringer-Ingelheim, Trovagene/Cardiff oncology, Incyte, Takeda, Novartis, Aprea, and ADC Therapeutics; participated in advisory boards, and/or had a consultancy with and received honoraria from AbbVie, Otsuka, Pfizer, Celgene/BMS, Jazz, Incyte, Agios, Boehringer-Ingelheim, Novartis, Acceleron, Astellas, Daiichi Sankyo, Cardinal Health, Taiho, Seattle Genetics, BeyondSpring, Trovagene/Cardiff Oncology, Takeda, Ionis, Amgen, Janssen, Epizyme, Syndax, Gilead, Kura, and Tyme; served on clinical trial committees for Novartis, Abbvie, Geron and Celgene/BMS; received travel support for meetings from Pfizer, Novartis, and Cardiff Oncology. **AD** has been a consultant for Bristol Myers Squibb, Novartis, Taiho, and Takenda. **UB** reports research funding from Takeda, Novartis, Jazz Pharmaceuticals, and Pfizer; membership on an entity's Board of Directors or advisory committees for Genentech, Daiichi Sankyo, Takeda, Novartis, and Pfizer; investigator in AbbVie-funded clinical trials. **GG-M** reports consultancy for Acceleron Pharmaceuticals, Astex Pharmaceuticals, Bristol-Myers Squibb, Celgene, Helsinn Therapeutics, Jazz Pharmaceuticals, and Genentech; honoraria for AbbVie, Acceleron Pharmaceuticals, Astex Pharmaceuticals, Celgene, and Helsinn Therapeutics; research funding from AbbVie, Amphivena Therapeutics, Astex Pharmaceuticals, Bristol-Myers Squibb, Celgene, Helsinn Therapeutics, H3 Biomedicine, Merck, Novartis, Genentech, and Onconova; membership on an entity's Board of Directors or advisory committees for Genentech. **IS, JRS, DP, PMR,** and **HS** are employees at Novartis. All other authors had nothing to disclose.

References

1. Platzbecker U, et al. *Leukemia*. Published online May 28, 2021. **2.** Lamarque M, et al. *Blood*. 2012;120(25):5084-5085. **3.** Pardoll DM. *Nat Rev Cancer*. 2012;12(4):252-264. **4.** Das M, et al. *Immunol Rev*. 2017;276(1):97-111. **5.** Kikushige Y, Miyamoto T. *Int J Hematol*. 2013;98(6):627-633. **6.** Kikushige Y, et al. *Cell Stem Cell*. 2010;7(6):708-717. **7.** Ngiow SF. *Cancer Res*. 2011;71(10):3540-3551. **8.** Sakuishi K, et al. *Trends Immunol*. 2011;32(8):345-349. **9.** Sabatos-Peyton C, et al. Oral presentation at *AACR 2016*. Session DDT01. **10.** Borate U, et al. Oral presentation at *ASH 2019*. Abstract 570. **11.** Acharya N, et al. *J Immunother Cancer*. 2020;8(1):e000911. **12.** Sabatos-Peyton C, et al. Poster presentation at *SITC 2020*. Abstract 439. **13.** Borate U, et al. Oral presentation at *EHA 2020*. Abstract S185. **14.** Wei AH, et al. Oral presentation at *EHA 2021*. Abstract S168. **15.** Zeidan AM, et al. Poster presentation at ASH 2020. **16.** Arber D, et al. *Blood.* 2016;127:2391-2405.