MDS 158

Updated Safety and Efficacy of Venetoclax in Combination With Azacitidine for the Treatment of Patients With Treatment-Naïve Higher-Risk Myelodysplastic Syndromes: Phase 1b Results

Andrew H Wei[1], Jacqueline S Garcia[2], Uma Borate[3], Chun Yew Fong[4], Maria R Baer[5], Daniel Nowak[6], Pierre Peterlin[7], Joseph Jurcic[8], Meagan A Jacoby[9], Uwe Platzbecker[10], Olatoyosi Odenike[11], Ilona Cunningham[12], Ying Zhou[13], David Hoffman[13], Leah Hogdal[13], Kiran Naqvi[14], Steve Kye[13], Guillermo Garcia-Manero[15]

[1]Department of Haematology, Alfred Hospital and Monash University, Melbourne, Victoria, Australia; [2]Department of Medical Oncology, Dana-Farber Cancer Institute, Boston, Massachusetts, United States; [3]Division of Hematology and Medical Oncology, Knight Cancer Institute, Oregon Health and Science University, Portland, Oregon, United States; [4]Olivia Newton John Cancer Research Institute, Austin Health, Melbourne, Victoria, Australia; [5]Greenebaum Comprehensive Cancer Center, University of Maryland School of Medicine, Baltimore, Maryland, United States; [6]Medical Faculty Mannheim of the Heidelberg University, Mannheim, Germany; [7]Nantes University Hospital, Nantes, France; [8]Herbert Irving Comprehensive Cancer Center, Columbia University Irving Medical Center, New York, New York, United States; [9]Siteman Cancer Center at Barnes-Jewish Hospital and Washington University School of Medicine, St Louis, Missouri, United States; [10]Medical Clinic and Policlinic 1, Hematology and Cellular Therapy, University Hospital Leipzig, Germany; [11]University of Chicago Medicine and Comprehensive Cancer Center, Chicago, Illinois, United States; [12]Haematology Department, Concord Repatriation General Hospital, University of Sydney, Sydney, New South Wales, Australia; [13]AbbVie Inc., North Chicago, Illinois, United States; [14]Genentech Inc., South San Francisco, California, United States; [15]Department of Leukemia, The University of Texas MD Anderson Cancer Center, Houston, Texas, United States

Presented at Society of Hematologic Oncology 2021 (SOHO) • September 8–11, 2021 • Houston, Texas

OBJECTIVE

- To update safety and efficacy findings of an ongoing, phase 1b study in patients with treatment-naïve higher-risk myelodysplastic syndromes (HR-MDS) treated with Venetoclax (Ven) + azacitidine (Aza)

BACKGROUND

- Hypomethylating agents (HMAs) are the current standard of care for patients with HR-MDS who are ineligible for allogeneic hematopoietic stem cell transplantation (HSCT)[1,2]
- Overall response rates (ORRs) remain suboptimal in patients receiving Aza, and their expected median overall survival (OS) is less than 2 years[3]
- Ven is a selective, potent, orally bioavailable B-cell lymphoma 2 (BCL-2) inhibitor, which has demonstrated synergy with HMAs in preclinical and clinical studies of myeloid malignancies[4–10]

Figure 1. Ven+Aza Mechanism of Action

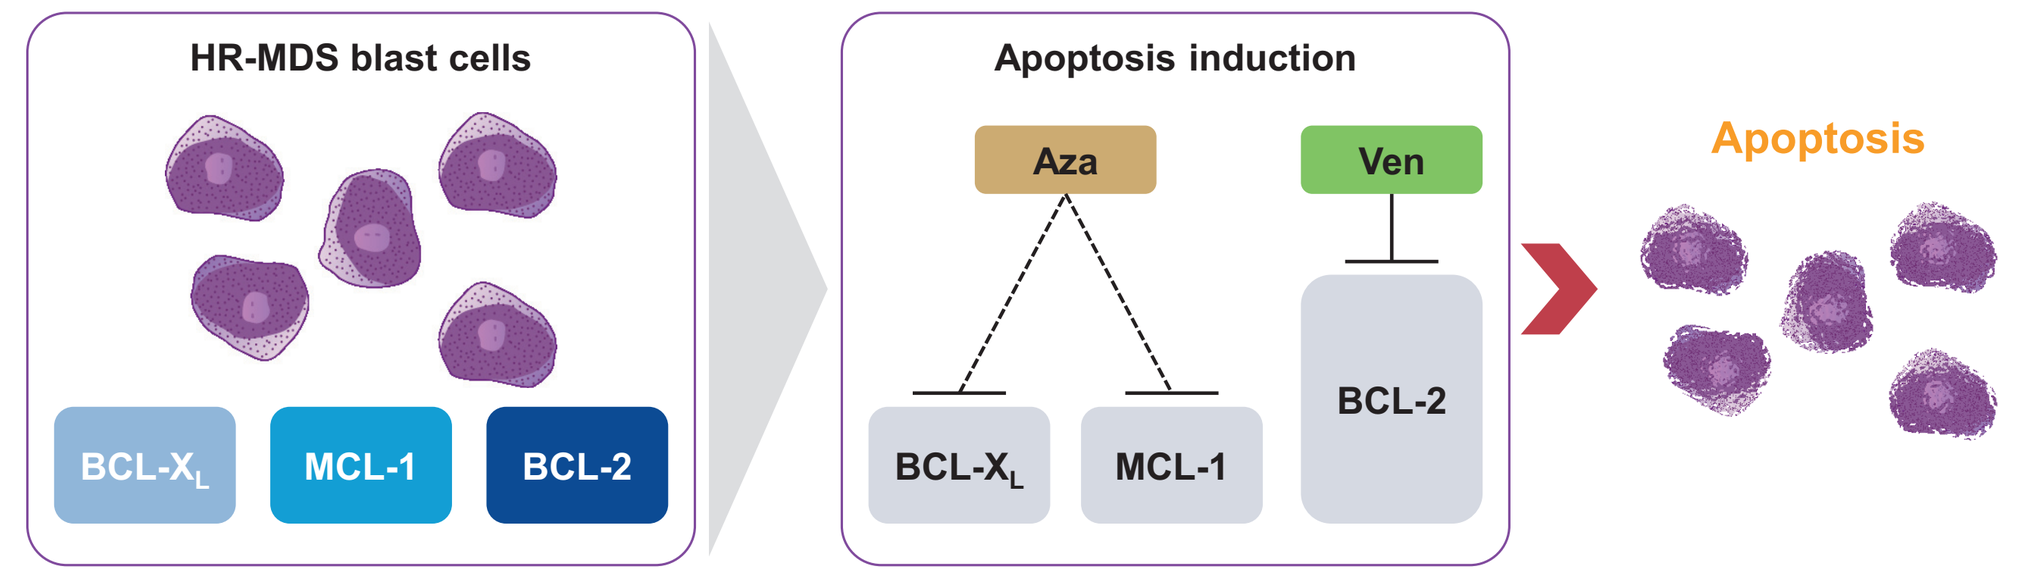
HR-MDS blast cells
BCL-XL
MCL-1
BCL-2
Apoptosis induction
Aza
Ven
BCL-XL
MCL-1
BCL-2
Apoptosis

Size of rectangles indicates relative dependency on specific protein for survival.
Dotted lines indicate an indirect therapeutic effect on BCL-2 family member dependency.

Aza, azacitidine; BCL, B-cell lymphoma; HR-MDS, higher-risk myelodysplastic syndromes; MCL, myeloid cell leukemia; Ven, venetoclax.
Jilg S, et al. *Exp Hematol Oncol*. 2019;8:9; Reidel V, et al. *Oncotarget*. 2018;9(25):17270–81; Jilg S, et al. *Leukemia*. 2016;30(1):112–23.

METHODS

Study Overview

M15-531 is an ongoing, phase 1b, dose-escalation, open-label, multicenter study evaluating Ven+Aza for the treatment of patients with HR-MDS (N = 78)

PRIMARY OBJECTIVES[a]

- Safety
- Establish the recommended phase 2 dose (RP2D)

KEY SECONDARY OBJECTIVES[a]

- Overall response rate (ORR)
- Overall survival (OS)

[a]Safety and efficacy assessments were carried out on all patients who received ≥1 dose of study drug; efficacy endpoints were evaluated per International Working Group 2006 response criteria.

KEY INCLUSION CRITERIA

- Adults ≥18 years
- No prior myelodysplastic syndrome (MDS) treatment
- International Prognostic Scoring System (IPSS) ≥1.5[a]
- Bone marrow blasts <20% at screening
- Eastern Cooperative Oncology Group (ECOG) score of ≤2

KEY EXCLUSION CRITERIA

- t-MDS, chronic myelomonocytic leukemia (CMML), u-MDS/MPN
- Patients planned to undergo intensive chemotherapy or allo-HSCT[b]
- CYP3A inducers within 7 days

[a]Study protocol has been amended to allow patients with higher-risk. [b]IPSS-Revised (intermediate, high, and very high) results and patients planning to undergo allo-HSCT, allogeneic hematopoietic stem cell transplantation.
MPN, myeloproliferative neoplasm.

METHODS (CONTINUED)

STUDY DESIGN

Treatment cohorts (28-day cycles); Aza 75 mg/m² D1–7.

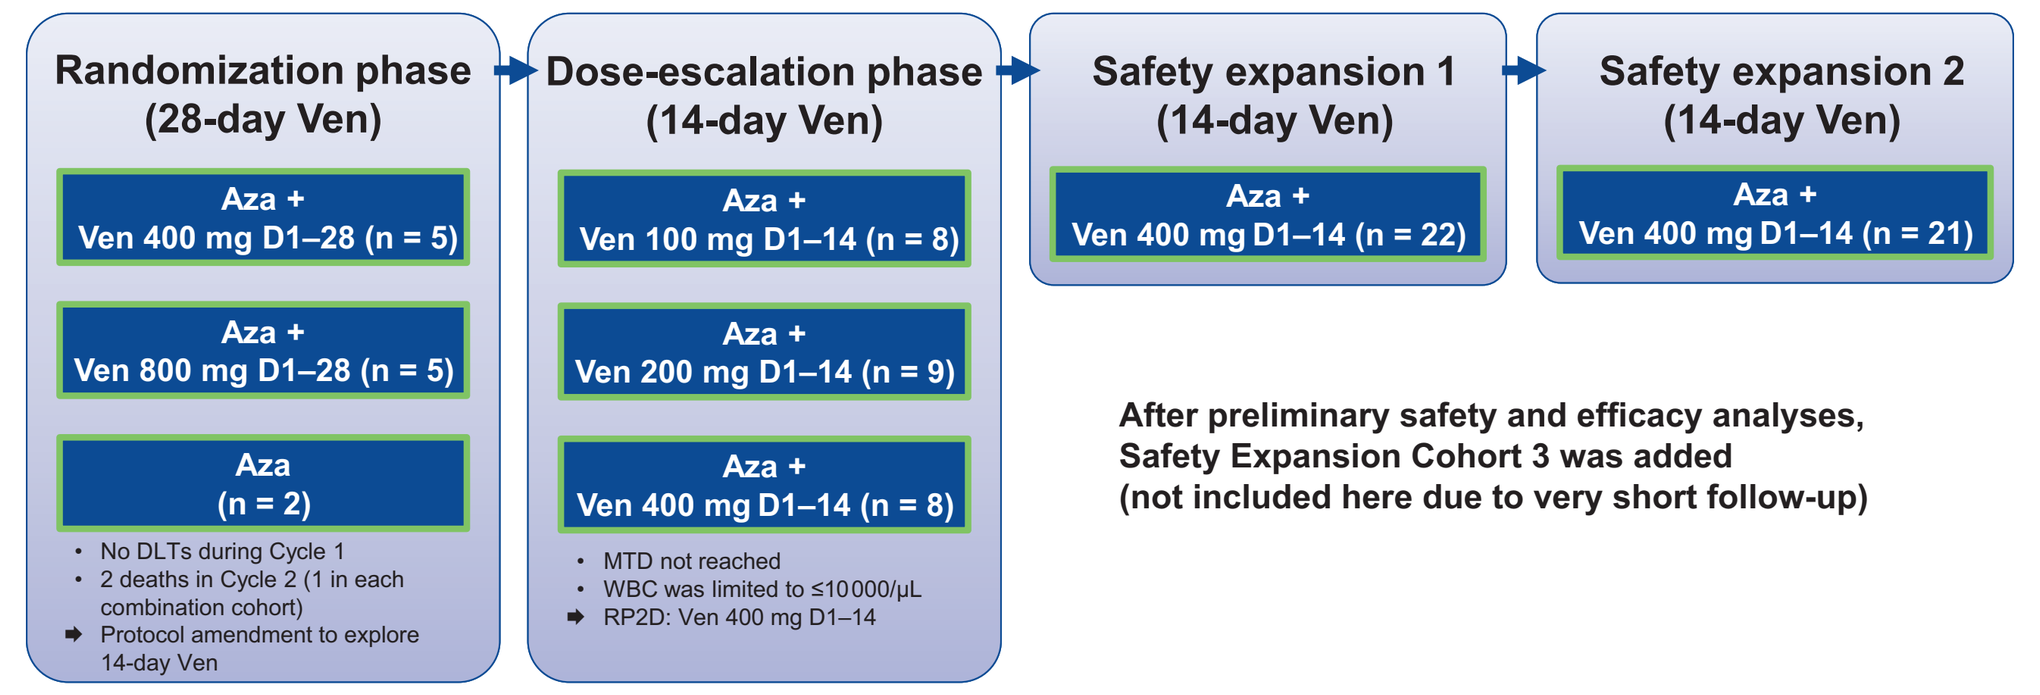
Randomization phase
(28-day Ven)
Aza + Ven 400 mg D1–28 (n = 5)
Aza + Ven 800 mg D1–28 (n = 5)
Aza (n = 2)
No DLTs during Cycle 1
2 deaths in Cycle 2 (1 in each combination cohort)
Protocol amendment to explore 14-day Ven
Dose-escalation phase
(14-day Ven)
Aza + Ven 100 mg D1–14 (n = 8)
Aza + Ven 200 mg D1–14 (n = 9)
Aza + Ven 400 mg D1–14 (n = 8)
MTD not reached
WBC was limited to ≤10000/μL
RP2D: Ven 400 mg D1–14
Safety expansion 1
(14-day Ven)
Aza + Ven 400 mg D1–14 (n = 22)
Safety expansion 2
(14-day Ven)
Aza + Ven 400 mg D1–14 (n = 21)
After preliminary safety and efficacy analyses, Safety Expansion Cohort 3 was added
(not included here due to very short follow-up)

Aza, azacitidine; D, Day; DLT, dose-limiting toxicity; MTD, maximum tolerated dose; RP2D, recommended phase 2 dose; Ven, venetoclax, WBC, white blood cell.

RESULTS

Median follow-up time was 23 months (range 0.1–44.2).

Table 1. Baseline Characteristics

Characteristic	n (% of N = 78)	
Male	56 (72)	
Median age, years [range]	70 [26–87]	
ECOG performance score		
0	33 (42)	
1	38 (49)	
2	7 (9)	
Bone marrow blasts		
≤5%	7 (9)	
>5% to ≤10%	21 (27)	
>10% to ≤20%	49 (63)	
>20%	1 (1)[a]	
IPSS karyotype risk		
Good	31 (40)	
Intermediate	16 (20)	
Poor	31 (40)	
IPSS risk classification		
Intermediate-2	57 (73)	
High	21 (27)	
IPSS-R risk classification		
Intermediate	14 (18)	
High	16 (20)	
Very high	48 (62)	
Baseline cytopenias	**grade 3**	**grade 4**
Neutropenia[b]	24 (31)	22 (28)
Thrombocytopenia[c]	20 (26)	6 (8)
Leukopenia[d]	30 (38)	3 (4)
Anemia[e]	10 (13)	0

[a]Patients recruited under 2017 Initial Protocol; [b]Includes neutrophil count decreased, febrile neutropenia, agranulocytosis, neutropenic sepsis, and neutropenic infection; [c]Includes platelet count decreased; [d]Includes white blood cell count decreased; [e]Includes hemoglobin count decreased. Data cutoff: Dec 15, 2020.
ECOG, Eastern Cooperative Oncology Group; IPSS, International Prognostic Scoring System; IPSS-R, International Prognostic Scoring System-Revised.

RESULTS (CONTINUED)

EXPOSURE, TOLERABILITY, AND SAFETY

- Median cycles of Aza received: 4 (range 1–27); median cycles of Ven received: 4 (range 1–27)
- 43 patients (55%) had >2 Ven dose intracycle interruptions
- Reasons for any dose interruption: non-hematologic AEs 58 (78%); hematologic toxicity 26 (35%); scheduling 21 (28%), other 42 (57%)
- 28 patients (36%) required ≥1 Ven dose reduction*
- Reasons for any Ven dose reductions: AEs 6 (21%); starting CYP3A inhibitor 21 (72%); other 7 (24%)
- 28 patients (36%) required ≥1 Aza dose reduction*
- 30-day mortality after first dose was 1%

*Dose reductions were typically 50% reduction for Ven and 33% reduction for Aza.

Table 2. Summary of Adverse Events

AEs, n (%)	Any grade	Grade 3	Grade 4
Neutropenia[a]	65 (83)	47 (60)	47 (60)
Febrile neutropenia	38 (49)	34 (44)	6 (8)
Nausea	43 (55)	1 (1)	0
Constipation	42 (54)	0	0
Diarrhea	38 (49)	2 (3)	0
Thrombocytopenia[b]	38 (49)	9 (11)	30 (38)
Vomiting	32 (41)	1 (1)	0
Leukopenia[c]	30 (38)	12 (15)	23 (29)
Anemia[d]	23 (29)	18 (23)	1 (1)
Fatigue	20 (26)	0	0
Hypokalemia	17 (22)	6 (8)	1 (1)
Dyspnea	16 (21)	1 (1)	0

Serious AEs, n (%)	57 (73)
Neutropenia[a]	38 (49)
Febrile neutropenia	35 (45)
Pneumonia	5 (6)
Sepsis	4 (5)
Diverticulitis	4 (5)

AEs reported in ≥20% of patients for any grade. SAEs reported in ≥5% of patients.
[a]Includes neutrophil count decreased, febrile neutropenia, agranulocytosis, neutropenic sepsis, and neutropenic infection; [b]Includes platelet count decreased; [c]Includes white blood cell count decreased; [d]Includes hemoglobin count decreased.
AEs, adverse events; Aza, azacytidine; SAEs, serious adverse events; Ven, venetoclax.

EFFICACY

23% of the study population moved to post-study allogeneic HSCT[e] prior to other post-study systemic cancer therapies.

Figure 3. Overall Response Rate

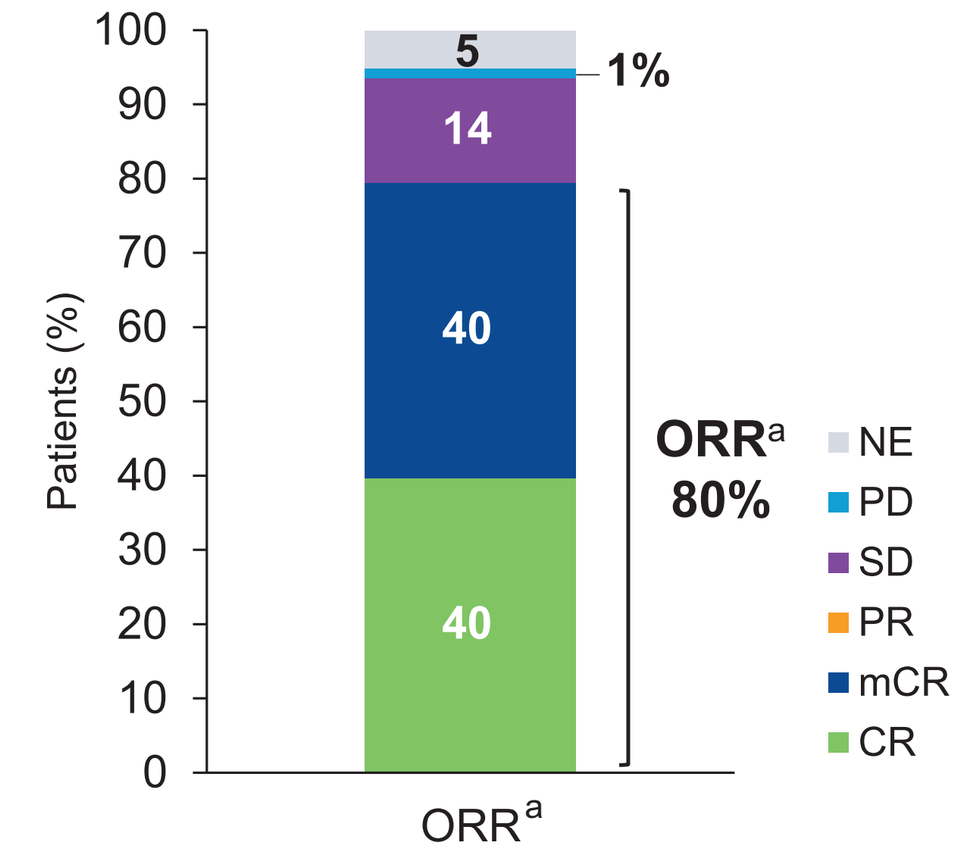
Patients (%)
100
90
80
70
60
50
40
30
20
10
0
5
1%
14
40
40
ORR[a]
80%
ORR[a]
NE
PD
SD
PR
mCR
CR

Table 3. Efficacy Summary

	n (% of N = 78)
Overall Response Rate[a]	62 (80%)
CR	31 (40%)
mCR	31 (40%)
mCR + hematologic improvement[b] n/N (%)	13/31 (42%)
Transfusion Independence Rate[c] n/N (%)	20/43 (46.5%)
[95% CI]	[31.2, 62.3]
	Months [95% CI]
Median time on study	23 [16.9, 24.5]
Overall Survival, median	28.2 [17.7, NE]
Overall Survival for CR, median[d]	28.6 [27.5, NE]
Duration of Response for CR, median	13.8 [8.9, NE]
Median time to CR, months (range)	2.6 (1.2–19.6)
Median time to mCR, months (range)	0.9 (0.7–4.6)

[a]ORR = CR+mCR+PR; PR n = 0; per IWG 2006; [b]Hematologic improvement (HI) includes patients who are eligible for HI evaluations and achieved neutrophil, erythroid, or platelet responses; [c]Defined as no transfusion ≥8 weeks; calculated for patients who were transfusion-dependent on RBC or platelet at baseline; [d]mOS for mCR+HI has not been reached; [e]Including bone marrow and peripheral blood stem cell.
CI, confidence interval; CR, complete remission; DoR, duration of response; mCR, marrow complete remission; NE, not estimable; ORR, overall response rate; PD, disease progression; PR, partial remission; SD, stable disease.

Figure 4. Clinical Activity Was Observed in Patients With HR-MDS Harboring Poor Prognostic Mutations

- Clinical responses were independent of baseline genetic mutation status
- ORR = CR+mCR+PR

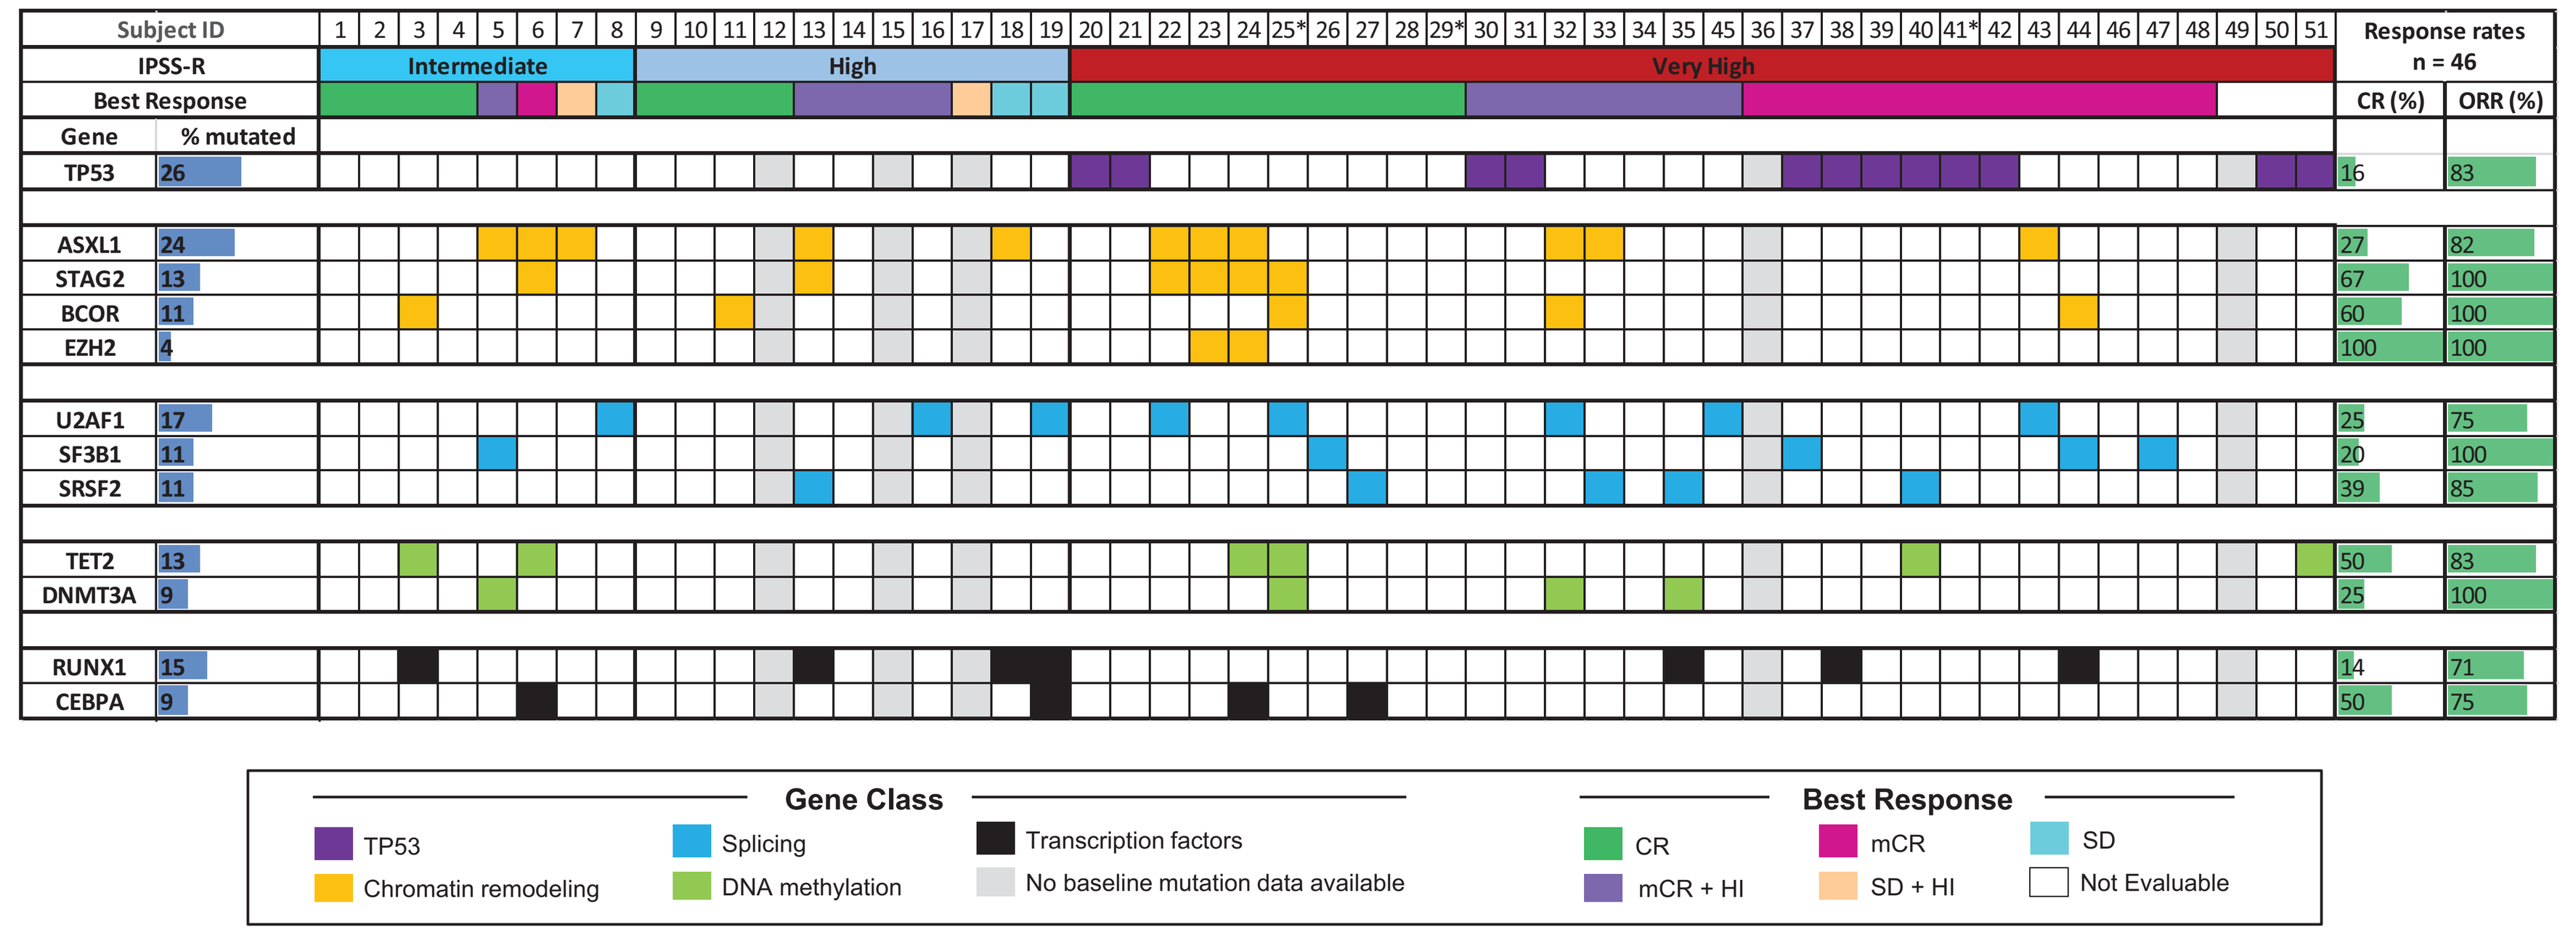
Subject ID
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25* 26 27 28 29* 30 31 32 33 34 35 45 36 37 38 39 40 41* 42 43 44 46 47 48 49 50 51
IPSS-R
Intermediate
High
Very High
Best Response
Gene
% mutated
Response rates
n = 46
CR (%)
ORR (%)
TP53 26 16 83
ASXL1 24 27 82
STAG2 13 67 100
BCOR 11 60 100
EZH2 4 100 100
U2AF1 17 25 75
SF3B1 11 20 100
SRSF2 11 39 85
TET2 13 50 83
DNMT3A 9 25 100
RUNX1 15 14 71
CEBPA 9 50 75
Gene Class
TP53
Chromatin remodeling
Splicing
DNA methylation
Transcription factors
No baseline mutation data available
Best Response
CR
mCR + HI
mCR
SD + HI
SD
Not Evaluable

Mutations assessed using BMA at screen using Archer Variant Plex Myeloid Gene Panel except for those with patients with * in which mutations were assessed using peripheral blood at SCR using Illumina TruSight Myeloid Gene Panel.
% mutated is calculated based on the 46 patients who have baseline mutations performed. Response rates based on IWG 2006 response criteria; Analysis of patients receiving RP2D and response rates are from Dec 2020 data cut.
CR, complete remission; HI, Hematologic Improvement; mCR, marrow complete remission; Mut, mutant; ORR, overall response rate; SD, stable disease.

Median time on the study: 23 months (range 0.1–44.2).

Figure 5. Overall Survival for All Patients

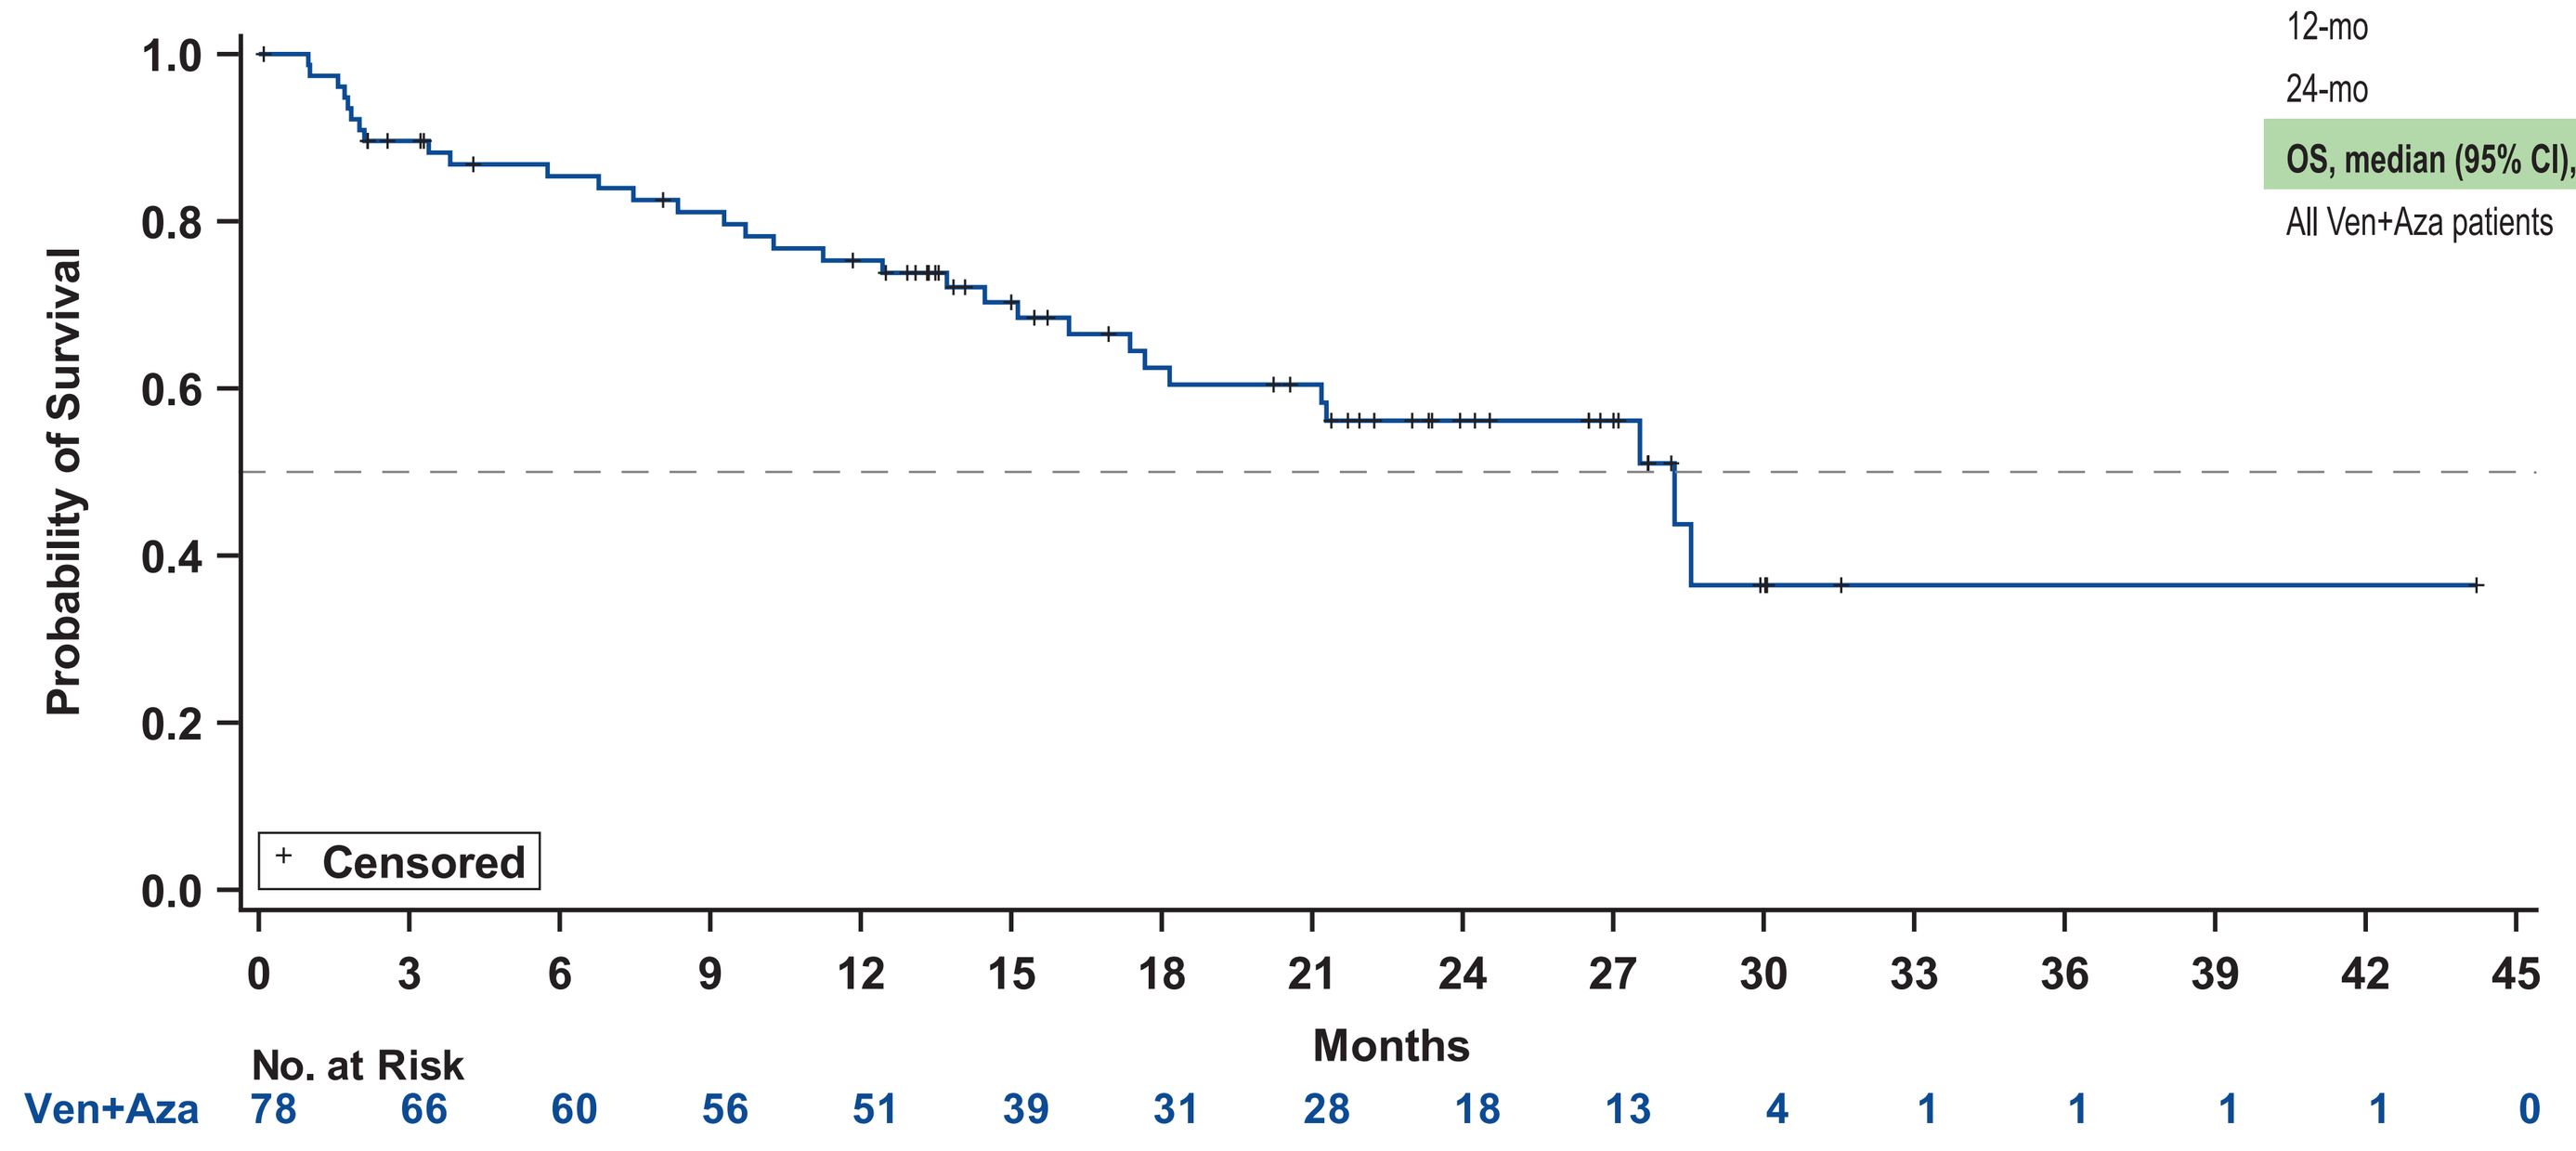
Probability of Survival
1.0
0.8
0.6
0.4
0.2
0.0
Censored
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45
Months
No. at Risk
Ven+Aza 78 66 60 56 51 39 31 28 18 13 4 1 1 1 1 0

Survival estimates, % (95% CI)	
12-mo	75.3 (63.6, 83.7)
24-mo	56.1 (42.5, 67.7)
OS, median (95% CI), months	
All Ven+Aza patients	28.2 (17.7, NR)

Aza, azacitidine; CI, confidence interval; NR, not reached; OS, overall survival; RP2D, recommended phase 2 dose; Ven, venetoclax.

Figure 6. Overall Survival by Response

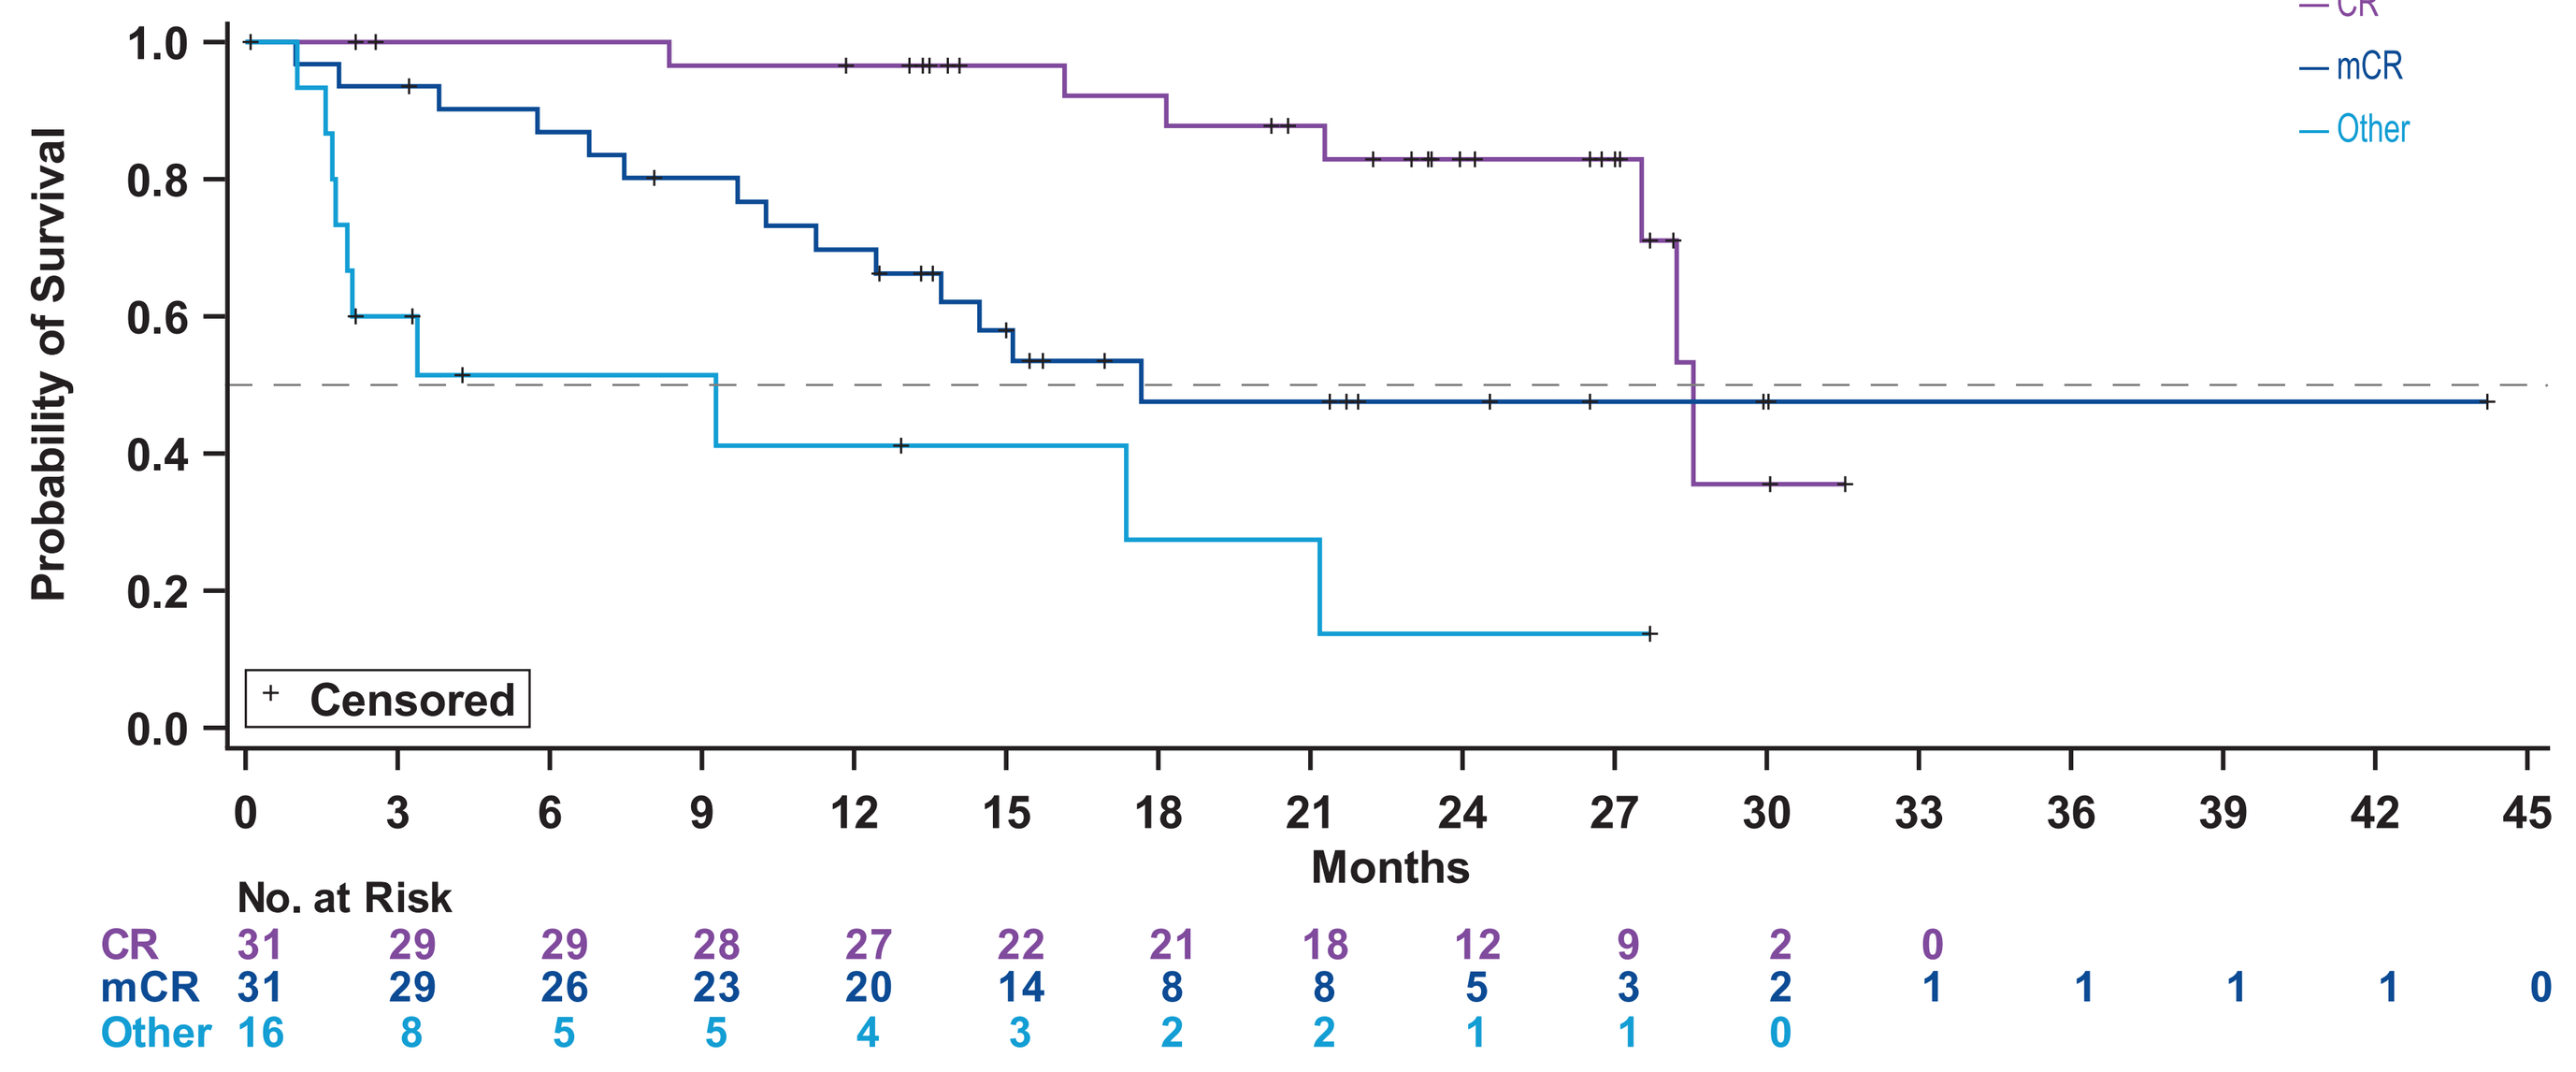
Probability of Survival
1.0
0.8
0.6
0.4
0.2
0.0
Censored
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45
Months
No. at Risk
CR 31 29 29 28 27 22 21 18 12 9 2 0
mCR 31 29 26 23 20 14 8 8 5 3 2 1 1 1 1 0
Other 16 8 5 5 4 3 2 2 1 1 0

OS, median (95% CI), months	
— CR	28.6 (27.5, NR)
— mCR	17.7 (11.3, NR)
— Other	9.3 (1.7, 21.2)

CI, confidence interval; CR, complete remission; mCR, marrow CR; NR, not reached; OS, overall survival.

CONCLUSIONS

- The combination of Ven+Aza was associated with a rapid and durable response, promising efficacy including remission rates and OS, and an acceptable safety profile for patients with HR-MDS
- Patients who achieved complete remission (CR) had median time to CR of 2.6 months and median duration of response was 13.8 months
- A Safety Expansion Cohort 3 was added, enrollment has concluded, and future analyses will include those data
- A phase 3 study (VERONA[a]) is currently recruiting patients to evaluate safety and efficacy in Ven+Aza in newly diagnosed patients with HR-MDS

[a]NCT04401748. Available at: https://clinicaltrials.gov/ct2/show/NCT04401748.

DISCLOSURES & ACKNOWLEDGMENTS

AH Wei: Honoraria: Novartis, Astellas, Pfizer, MacroGenics, AbbVie, Genentech, Servier, Celgene, Amgen, AstraZeneca, Janssen. Research funding: Novartis, Celgene, AbbVie, Servier, AstraZeneca, and Amgen. Former employee of the Walter and Eliza Hall Institute and receives a fraction of its royalty stream related to venetoclax. **JS Garcia:** Research support from AbbVie. Clinical trial support (institutional) from AbbVie, Genentech, AstraZeneca, Prelude, and Pfizer. Advisory board for: AbbVie, Astellas, and Takeda. **U Borate:** Advisory board: Genentech, Daiichi-Sankyo, Takeda, Pfizer, Novartis. Research funding: Novartis, Pfizer, Jazz Pharma. Consultant: AbbVie. Investigator: AbbVie-funded clinical trials. **CY Fong:** Honoraria: AbbVie, Amgen, Novartis, Pfizer, Astellas. Research funding: Amgen. Speakers bureau: Amgen, BMS. **MR Baer:** Research funding (institutional): AbbVie, Astellas, Forma, Incyte, Kite, Oscotec, Takeda. **D Nowak:** Research funding: Tolero Pharma, Pharmaxis, Apogenix. Investigator: AbbVie-funded clinical trial. **P Peterlin:** Honoraria: Astellas, Jazz Pharma, AbbVie. Research funding: Jazz Pharma, and Astellas. **J Jurcic:** Consulting or advisory role: AbbVie, Celgene, Daiichi-Sankyo, Novartis. Research funding (institutional): AbbVie, Arog Pharmaceuticals, Astellas, Celgene, Daiichi-Sankyo, Forma Therapeutics, Genentech, Kura Oncology, PTC Therapeutics, Syros Pharmaceuticals. **MA Jacoby:** Research funding: AbbVie, Jazz Pharmaceuticals. **U Platzbecker:** Data monitoring committee: Onconova, Takeda, Novartis, Celgene/BMS. Consulting: Celgene/BMS, Novartis, BMS, Daiichi-Sankyo, Agios. Research support from AbbVie. **O Odenike:** Consulting or advisory role: AbbVie, Celgene, Impact Biomedicines, Novartis, Taiho Oncology, Takeda Pharmaceutical. Research funding: Celgene (Inst), Incyte (Inst), AstraZeneca, Astex Pharmaceuticals (Inst), NS Pharma (Inst), AbbVie (Inst), Gilead Sciences (Inst), Janssen Oncology (Inst), Oncotherapy (Inst), Agios (Inst), CTI/Baxalta (Inst), Aprea (Inst). **I Cunningham:** Research funding: AbbVie, Amgen, Astex, Celgene, Janssen, Novartis, Principia Biopharma, Rigel. **Y Zhou, D Hoffman, L Hogdal,** and **S Kye:** Employees of AbbVie and may own stock. **K Naqvi:** Employee of Genentech and may own Roche stock or stock options. **G Garcia-Manero:** No conflicts, Investigator in AbbVie-funded Clinical Trials.

Venetoclax is being developed in a collaboration between AbbVie and Genentech. AbbVie and Genentech provided financial support for the study and participated in the design, study conduct, analysis, and interpretation of data, as well as the writing, review, and approval of the abstract. No honoraria or payments were made for authorship.

The authors, AbbVie, and Genentech would like to thank the patients, their families and caregivers, as well as the study investigators, research, and supporting staff. Medical writing and support was provided by Ana Mrejeru, PhD (AbbVie employee).

REFERENCES

1. NCCN Guidelines®. Myelodysplastic Syndromes. V3.2021.
2. Fenaux P, et al. *Lancet Oncol*. 2009;10(3):223–32.
3. Sekeres MA, et al. *J Clin Oncol*. 2017;35(24):2745–53.
4. DiNardo CD, et al. *Lancet Oncol*. 2018;19(2):216–28.
5. Wei AH, et al. *J Clin Oncol*. 2019;37(15):1277–84.
6. DiNardo CD, et al. *Am J Hematol*. 2018;93(3):401–7.
7. Jilg S, et al. *Exp Hematol Oncol*. 2019;8:9.
8. Jin S, et al. *Clin Cancer Res*. 2020;26(13):3371–83.
9. Reidel V, et al. *Oncotarget*. 2018;9(25):17270–81.
10. Wei AH, et al. *Blood*. 2020;135(24):2137–45

Scan QR code to download an electronic version of this poster (MDS 158) and other AbbVie SOHO 2021 Scientific Presentations

QR code expiration: August 8, 2022.

To submit a medical question, please visit www.abbviemedinfo.com.

© AbbVie Inc. 2021