MDS-082

Pauline Frank, pauline.frank@novartis.com

Experiences and Support Needs of Caregivers of Patients With Higher-Risk Myelodysplastic Syndrome: Qualitative Insights via Online Bulletin Board (OBB) in the US

Pauline Frank[1], Anne Olshan[2], Tracey Iraca[3], Cindy Anthony[4], Sophie Wintrich[5], Emma Sasse[1]

[1]Novartis Pharma AG, Basel, Switzerland; [2]Olshan Patient Relations Inc, Stamford, CT, USA; [3]Myelodysplastic Syndromes Foundation US, Yardville, NJ, USA; [4]Aplastic Anemia & Myelodysplasia Association Canada, ON, Canada; [5]MDS UK Patient Support Group, London, UK

Scan to obtain:
- Poster
- Supplementary Podcast

http://novartis.medicalcongressposters.com/Default.aspx?doc=ab968

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

CONCLUSIONS

- The findings highlight a significant impact and burden on caregivers of patients with higher-risk MDS, which varies depending on the stage of the disease, the choice (or lack of choice) of treatments, and the personal situation of the caregiver
- For example, although the caregiver role for more recently diagnosed patients requires limited efforts, their role increases significantly if a stem cell transplant occurs and/or if the patient's health status changes
- Emotional stress is pervasive, especially at the diagnosis and prognosis stage and when told to "watch and wait"
- This is amplified when patients/caregivers perceive a lack of knowledge/expertise about MDS in HCPs, resulting in higher burden as they seek information and additional expert advice
- Additional stress factors include the financial burden, time burden when balancing other commitments, and impact on their own health—particularly if they have their own health issues to manage
- Most caregivers can manage the physical and functional aspects of care; however, many state that the bigger unmet need for both patient and caregiver is emotional support, which has not typically been part of the standard of care provided to patients with MDS
- There is a strong need for mental health support for both patient and caregiver
- There is an opportunity for healthcare systems to provide better support in these areas of emotional burden

This study is sponsored by Novartis Pharmaceuticals Corporation.
Presented at the Society of Hematologic Oncology Ninth Hybrid Annual Meeting; September 8-11, 2021.

INTRODUCTION

- Myelodysplastic syndromes (MDS) are a heterogeneous group of hematopoietic diseases that are associated with impaired bone marrow function, persistent cytopenia, and a risk of progression to acute myeloid leukemia (AML)[1]
- Patients with MDS, especially those with higher-risk MDS, face considerable challenges in the course of disease management and often require support from a caregiver
- Patients with advanced cancer and their caregivers can experience physical, emotional, social, financial, and functional issues
- Research on the burden of caring for patients with advanced cancer suggests that caregivers receive insufficient support, likely due to the lack of knowledge about their needs[2-4]
- Prospective evidence shows that having the consistent support of an inpatient care-partner is associated with longer survival in patients receiving a bone marrow transplant[5]
- Although not yet studied in the area of higher-risk MDS, these findings give weight to a strategy to support both patient and caregiver for potential better outcomes in MDS
- The overall experience of caregivers for patients with MDS has not been evaluated to date

OBJECTIVES

- To identify key challenges for caregivers of patients with higher-risk MDS before, during, and after diagnosis and along the disease journey; impact on the caregiver's life and well-being; and impact of decisions made, including treatments

METHODS

Qualitative Research Design

- The OBB was conducted over 14 days and enabled data collection via a web-based platform
- A mix of moderator-led question-and-answer sessions and participant discussion provided the caregiver insights as online dialogue
- This type of online focus group can generate more meaningful and impactful insights compared with traditional in-person focus groups[6]

Participants

- Eligible participants were aged ≥18 years, caregivers of an adult patient with high- or very high-risk MDS or intermediate-risk patients receiving hypomethylating agents, and proficient in written English
- Participants were excluded if they were a caregiver of a patient with lower-risk MDS, a patient in a clinical trial, or a paid caregiver

Analyses

- A content analysis of the full text was conducted independently by 2 researchers per country (United States, Canada, and United Kingdom)
- The findings were compared across countries to identify common themes and categories
- The results from the US caregivers are presented in this poster

RESULTS

Participants

- 5 caregivers were from the United States—all were White and women (**Figure 1**)
- Care was provided for patients in their 60th (n=1), 70th (n=3), and 80th (n=1) decile of life

Figure 1. Caregiver Age and Education Level

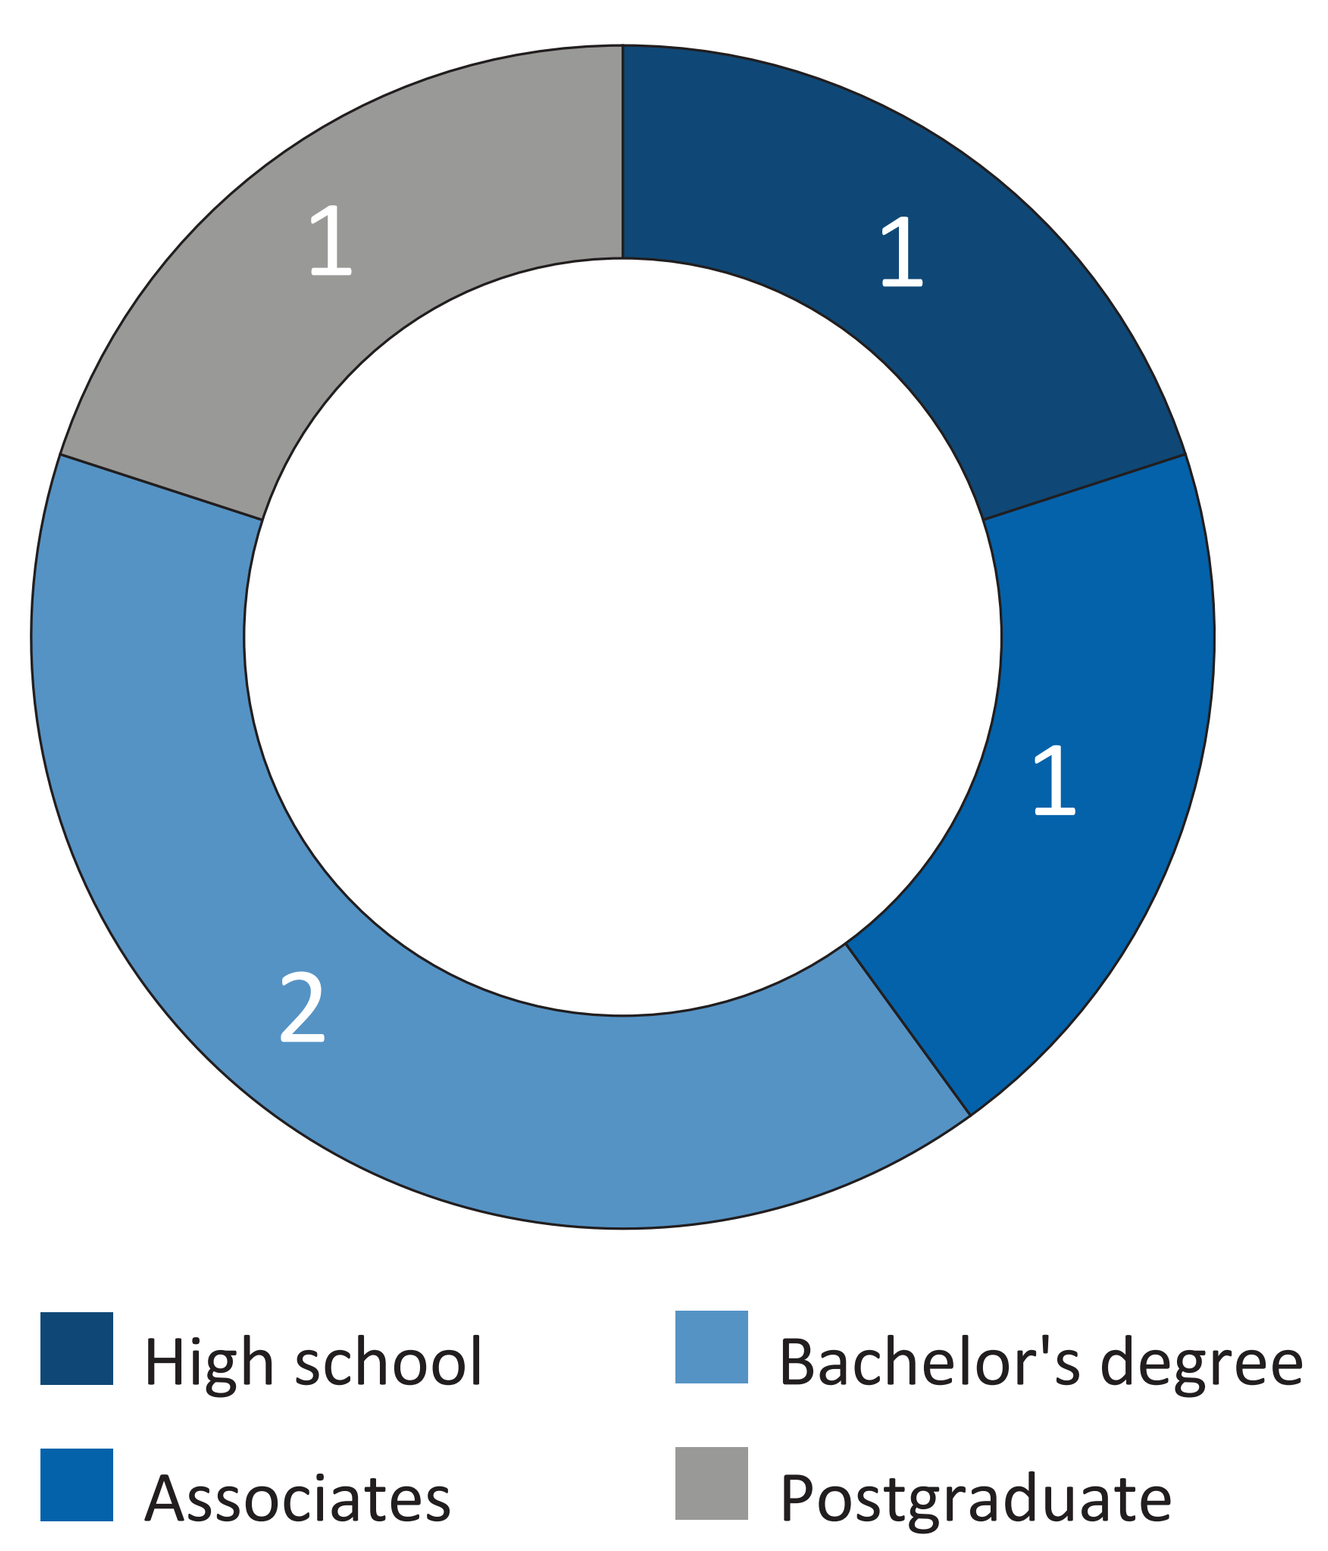
Caregiver Education Level
1
1
1
2
High school
Bachelor's degree
Associates
Postgraduate

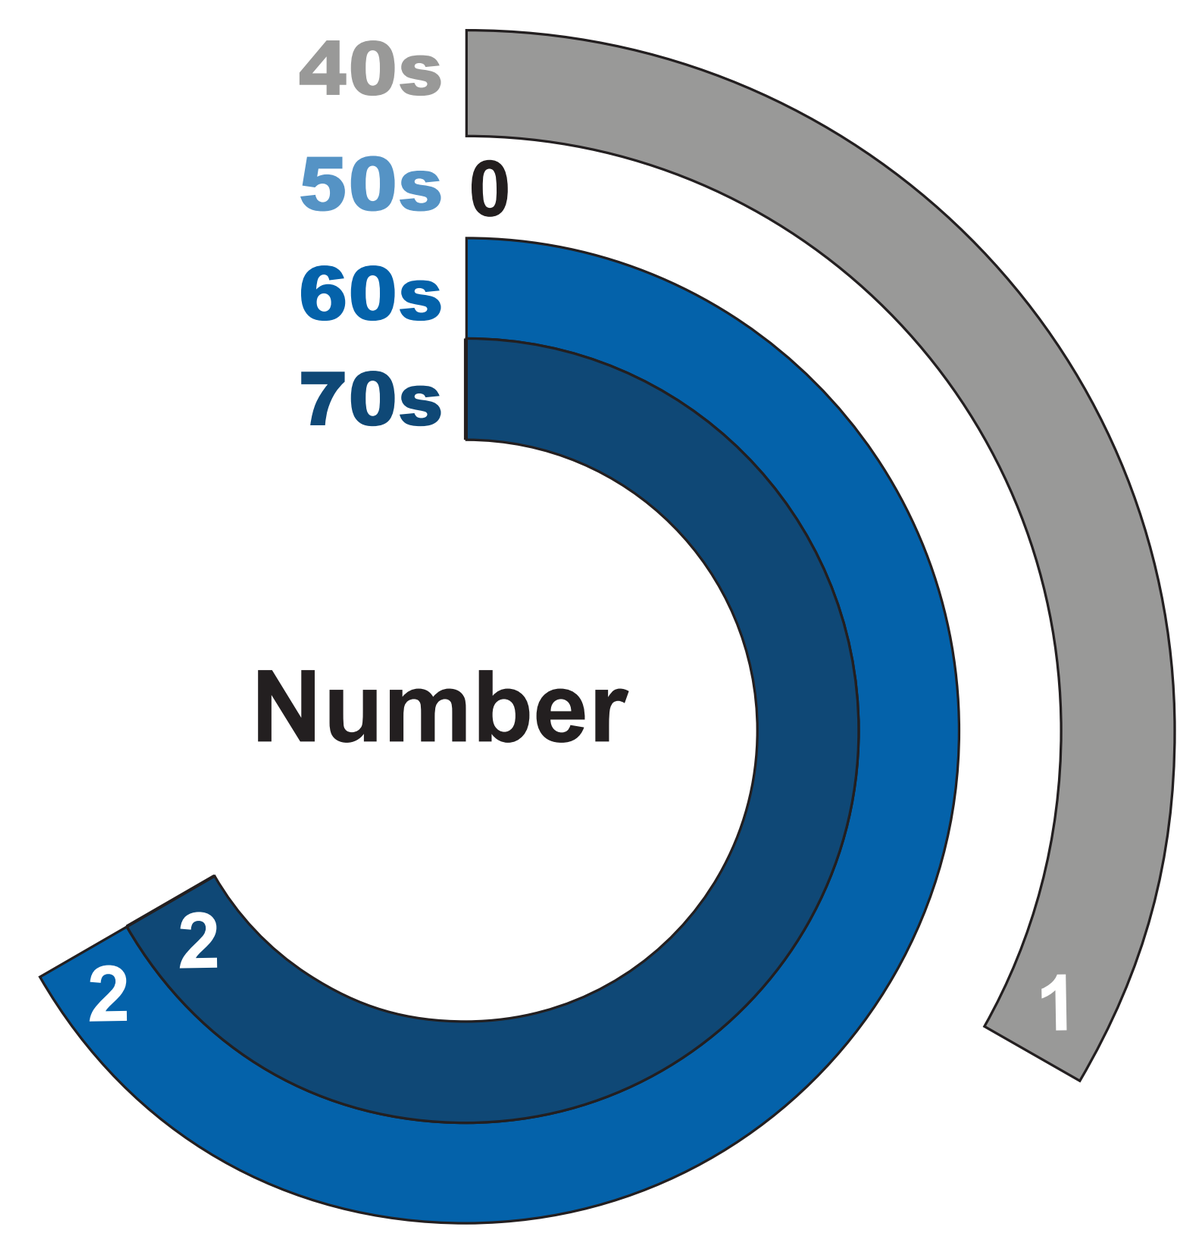
Caregiver Age Decile, years
40s
50s 0
60s
70s
Number
2
2
1

- Findings from the OBB data revealed the strong impact and burden on caregivers of patients with higher risk-MDS, which varies depending on disease stage, the choice of treatments, and the caregiver's personal situation
- 7 key themes were identified where the caregiver burden was significant

Key Themes Experienced by Caregivers of Patients With Intermediate- or Higher-Risk MDS

"I was solely responsible. I was [my husband's] advocate and being assertive to doctors and nurses was not something I was looking forward to, but I had more strength than I knew."

HCP interactions
- Common sources of dissatisfaction with HCPs include poor or insufficient communication about disease complexities, hesitancy to share prognosis, limited treatment options, and consideration of family needs and requests
- Lack of a multidisciplinary approach is disruptive to patient-HCP communication
- Caregivers may prefer MDS Centers of Excellence even when factoring in the increased burdens on time, appointment logistics, etc

Treatment experience
- Limited treatment options bring unexpected burden (eg, transplant) or loss of hope with limited control of the disease (eg, hypomethylating agents)
- As patient physically and/or mentally declines, role of "decision-maker" increases for caregiver and is aided by seeking information on the internet and/or a second opinion

Mental health
- Emotional stress is pervasive throughout the disease journey, with peaks in several phases: at diagnosis, when making treatment choices, and when treatment fails
- Caregiver is protective of the patient and absorbs the physical, functional, and emotional needs of the patient—often acting as decision-maker and motivator; however, they do not share their own needs or negative feelings with the patient
- Often feels alone, experiencing a lack of empathy from others
- Feels that life has been limited, with lowered future expectations; future plans put on hold as life now revolves around the patient's needs
- Coping mechanisms include lowering life expectations, having faith, and making time for themselves
- Contact with family/friends (and/or other patients/caregivers in-person or online) is an important social support network

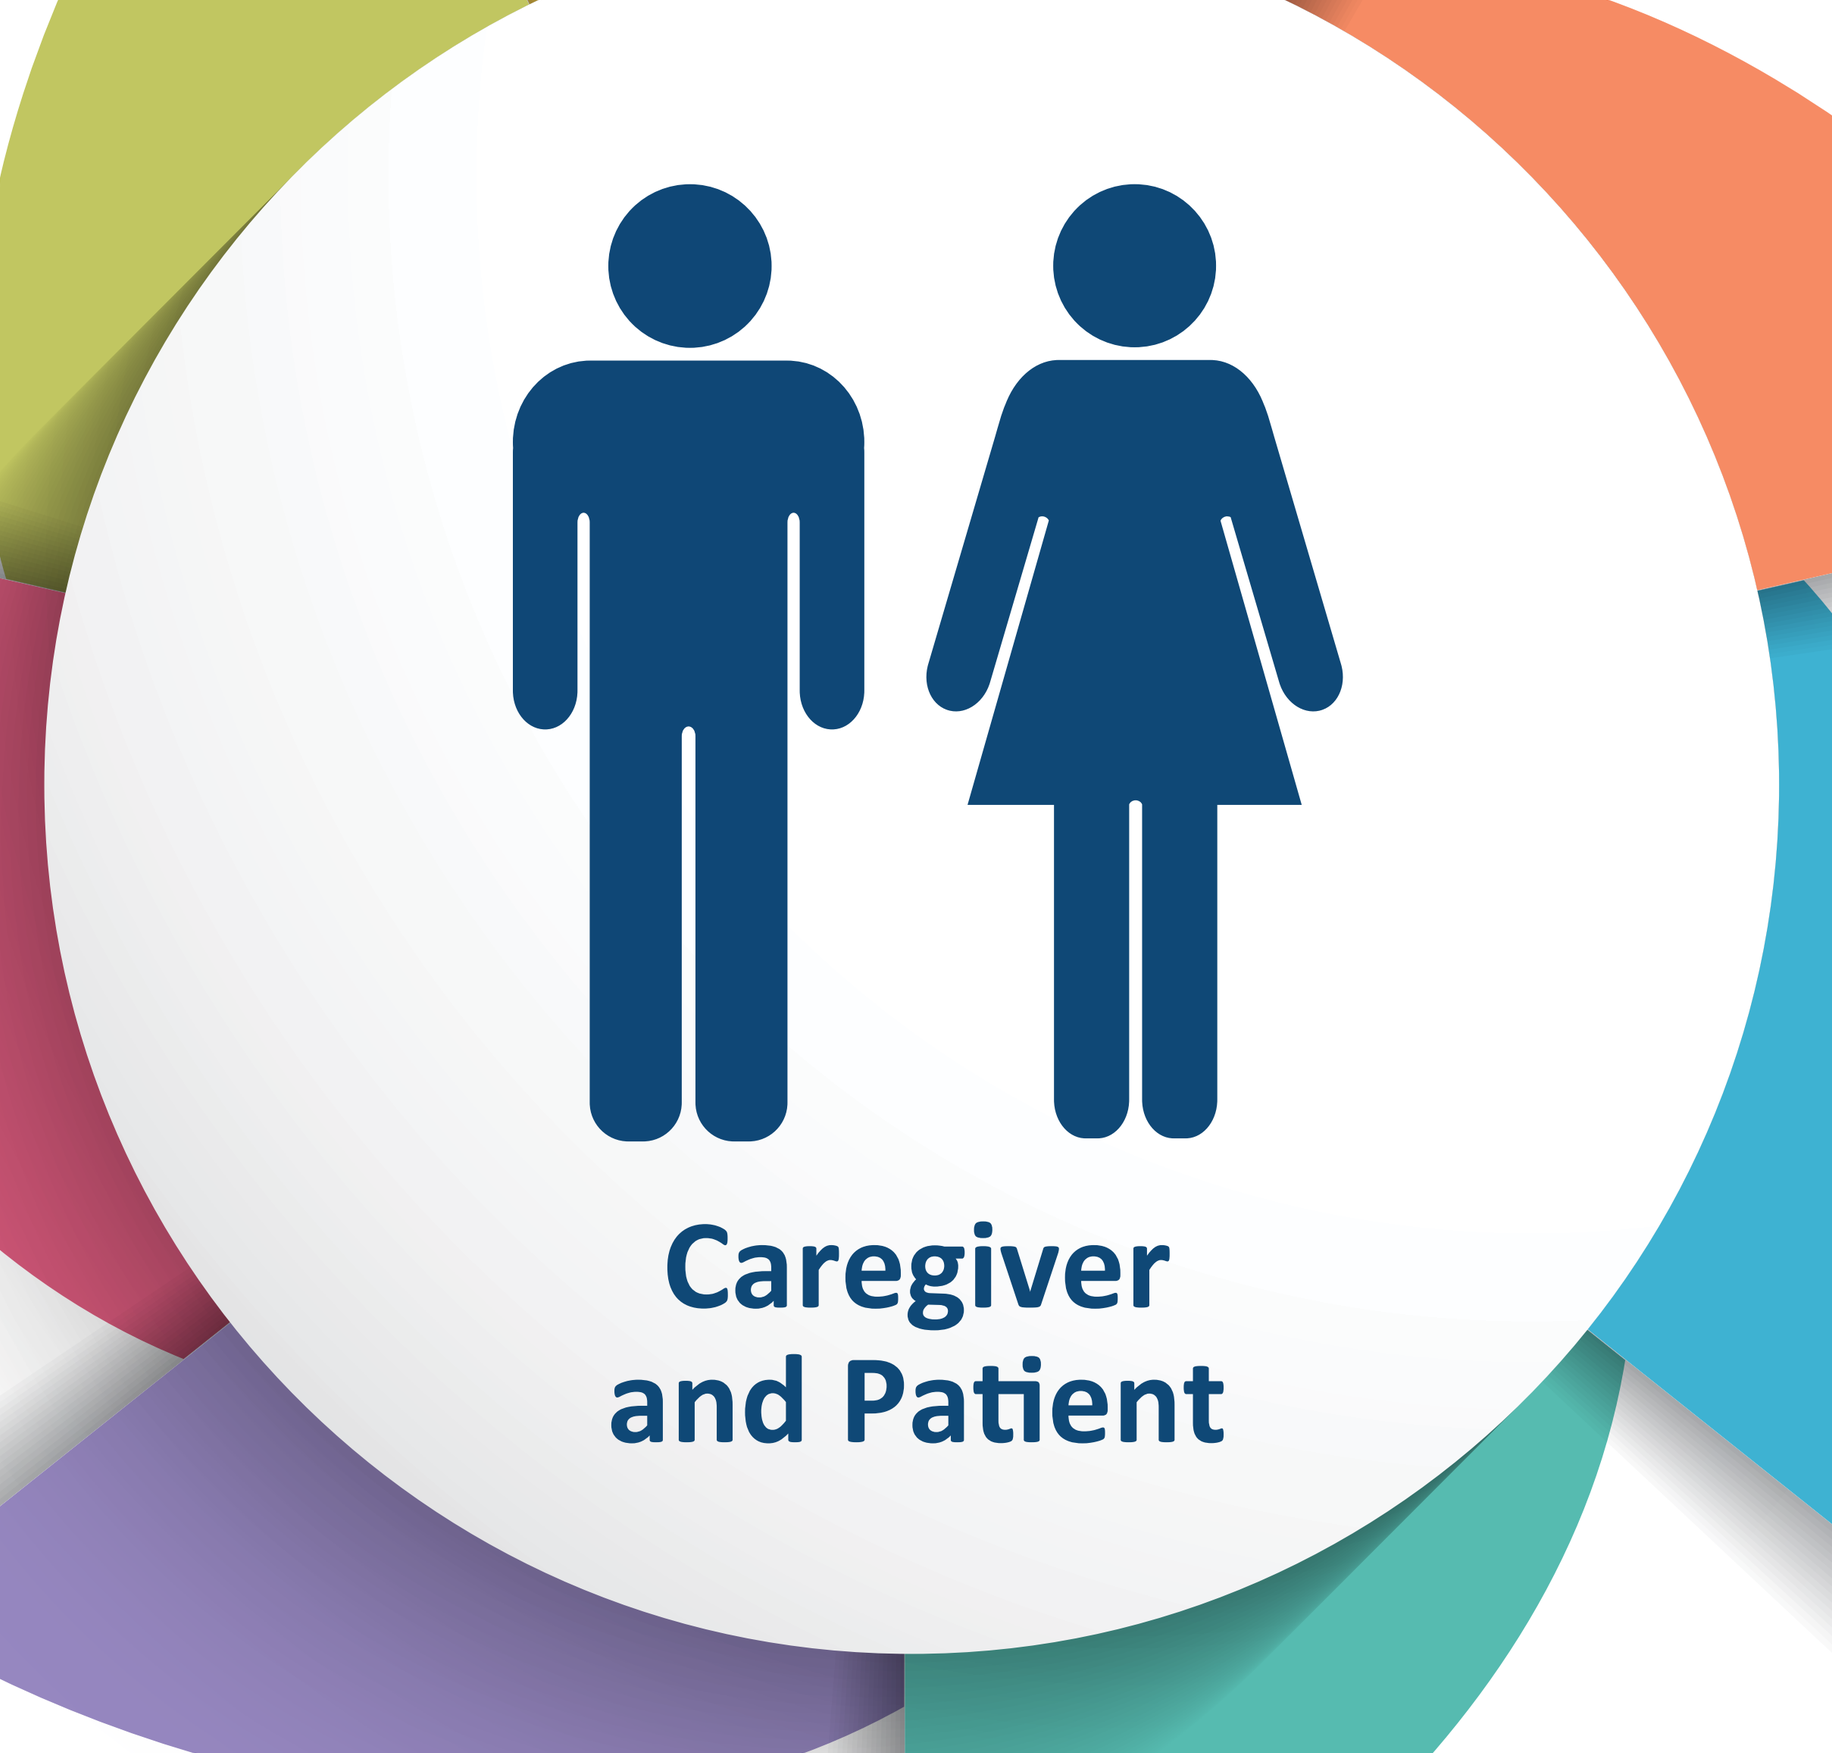
Caregiver and Patient

Caregiver role & burden
- Partner caregiver puts patients' needs first (including own health care issues)
- Children caregivers adapt their family/work lives to support the patient on information, logistics, and access to treatments
- Level and burden of care changes as the disease progresses and/or depending on intensity of treatment; transplant recipients require substantially more care
- The caregiver burden is perceived to be less in the earlier years of care (vs other countries with longer years of caregiver experience)
- Financial impact is increased but manageable (vs other countries with greater impact on caregiver employment status and financial stress)

"We moved to a different state in order to be close to a Center of Excellence. I knew no one and my whole support system was gone."

Family dynamics
- Motivating the patient to be more active can be perceived as being nagging/bossy
- May choose to avoid having conversations regarding prognosis or treatment failure to protect patient and keep them hopeful
- Most difficult situations arise when patient and caregiver are not communicating well
- Co-caregiving benefits include clear roles for each caregiver and less impact on each person's lives
- Differences in perspectives/information can bring conflict among co-caregivers

"[My husband is] too tired or he doesn't want to think about the hard questions that the doctor will be asking him."

"In my darkest times I called [the] MDS [Foundation] for support. Later after the transplant I called BeTheMatch for caregiver support. There you get matched with another caregiver who has had a similar experience. That was extremely helpful. At the hospital we also talked to the social worker and to volunteers who had had a transplant. That was always very emotional but very helpful."

Information & education
- Decision-making is challenging because of the disease complexity and perceived inaccurate/incomplete information
- Trust is undermined by a lack of consensus among different physicians regarding new treatments
- Patient advocacy groups are perceived as a source of reliable and clear information
- Social media is perceived as a positive environment in which to share knowledge and experience with other patients/caregivers

Disease experience
- Lack of awareness about MDS, its complexity, its poor prognosis, and its limited curative treatment options contribute to fear, uncertainty, and loss of hope
- Feelings can be compounded by poor communication by HCP, leading to a sense of paralysis for some; this situation can also motivate others to seek information and a second opinion
- The "watch-and-wait" approach is stressful and difficult to understand; receiving active treatment, even with side effects, is seen as a blessing in comparison
- Patient quality of life and survival are main goals for the caregiver

HCP, healthcare provider.

Acknowledgements

The authors thank the caregivers who participated in the research and their families.

Financial support for medical editorial assistance was provided by Novartis Pharmaceuticals Corporation. The authors thank Apurva Davé, PhD, of Medical Expressions, for medical editorial assistance with this poster presentation.

Disclosures

All authors had nothing to disclose.

References

1. Platzbecker U, et al. *Leukemia*. 2021;35(8):2182-2198.
2. Chambers SK, et al. *Eur J Cancer Care* (Engl). 2012;21(2):213-223.
3. Friðriksdóttir N, et al. *Acta Oncol*. 2011;50(2):252-258.
4. Sklenarova H, et al. *Cancer*. 2015;121(9):1513-1519.
5. Foster LW, et al. *Bone Marrow Transplant*. 2013;48(5):671-677.
6. Reid D, Reid F. *Int J Market Res*. 2005:47:131-162.