Assessment of Fixed-Duration Therapies for Treatment-Naïve Waldenström Macroglobulinemia

Jithma P. Abeykoon, M.D.[1], Saurabh Zanwar, M.D.[1], Stephen M. Ansell, M.D., Ph.D.[1], Eli Muchtar, M.D.[1], Rong He, M.D.[2], Patricia T. Greipp, M.D.[3], Rebecca L. King, M.D.[2], Sikander Ailawadhi, M.D.[4], Jonas Paludo, M.D.[1], Jeremy T. Larsen, M.D.[5], Thomas M. Habermann, M.D.[1], David Inwards, M.D.[1], Ronald S. Go, M.D.[1], Gita Thanarajasingam, M.D.[1], Francis Buadi, M.D.[1], Angela Dispenzieri, M.D.[1], Carrie A. Thompson, M.D.[1], Thomas E. Witzig, M.D.[1], Martha Lacy, M.D.[1], Wilson Gonsalves, M.D.[1], Grzegorz S. Nowakowski, M.D.[1], David Dingli, M.D., Ph.D[1], S. Vincent Rajkumar, M.D.[1], Robert A. Kyle, M.D.[1], Taimur Sher, M.D.[4], Vivek Roy, M.D.[4], Allison Rosenthal, M.D.[5], Asher A. Chanan-Khan, M.D.[4], Craig Reeder, M.D.[5], Morie A. Gertz, M.D.[1], Shaji Kumar, M.D.[1], and Prashant Kapoor, M.D.[1]

[1]Division of Hematology; [2]Department of Laboratory Medicine and Pathology; [3]Division of Laboratory Genetics and Genomics; Mayo Clinic, Rochester, Minnesota

[4]Division of Hematology and Oncology, Mayo Clinic, Florida, [5]Division of Hematology, Mayo Clinic, Arizona

INTRODUCTION

- Waldenström macroglobulinemia (WM) is an indolent, B-cell non-Hodgkin lymphoma associated with monoclonal IgM circulating protein and lymphoplasmacytic marrow infiltration.
- The anti-CD20 monoclonal antibody, rituximab is the cornerstone of frontline therapy of WM and used frequently as monotherapy or in combination with alkylating agents, proteasome inhibitors (PI) or ibrutinib.
- A substantial proportion of noteworthy recent clinical trials in this rare malignancy have combined both previously untreated and relapsed and/or refractory WM patient populations and are not necessarily adequately powered to discern the impact of initial therapies in different clinical settings.
- Therefore, we attempted to compare the efficacy of the most frequently used rituximab-based fixed-duration anti-WM regimens in routine practice [rituximab plus bendamustine (R-Benda); dexamethasone, rituximab, cyclophosphamide (DRC); and bortezomib, dexamethasone, rituximab (BDR)] to inform clinicians about the benefits and limitations of choosing one regimen over the other.

METHODS

DESIGN: A retrospective cohort study.

PARTICIPANTS: Patients with active WM seen at Mayo Clinic Rochester, Arizona and Florida campuses between 10/01/2000 and 10/31/2019 who received R-Benda, DRC, or BDR as primary therapy were included.

OUTCOMES MEASURES: Overall response rate (ORR), event-free survival (EFS), progression-free survival (PFS), overall survival (OS), time-to-next therapy (TTNT) and treatment related adverse-effects (AEs) based on the three treatment regimens. The *MYD88* ^{L265P} mutation status was assessed using allele-specific polymerase-chain-reaction (sensitivity 1%).

Response rates were assessed by modified Sixth International Workshop on WM Criteria. Time-to-event analyses were performed from the frontline therapy, using Kaplan-Meier method. CTCAE v4.03 was used to grade toxicities

TREATMENT:

- Bendamustine in R-Benda was given at 90 mg/m2 intravenously (IV) on days 1 and 2 (with dose reduction to 70mg/m2 for the elderly population) and rituximab at 375 mg/m2 intravenously on day 1 of the 28-day cycle.
- The DRC regimen consisted of 21-day courses of dexamethasone IV 20 mg, rituximab IV 375 mg/m2 on day 1 and oral cyclophosphamide 100 mg/m2 orally twice daily on days 1 to 5.
- In the BDR regimen, given for upto 23 weeks, the duration of Cycle 1 was 21 days, while Cycles 2 to 5 were built for 35 days per cycle. Bortezomib was given at 1.3 mg/m2 IV or subcutaneously (SQ) on day 1, 4, 8, and 11 of Cycle 1, and at 1.6 mg/m2 in subsequent cycles with dexamethasone at 40 mg on days 1, 8, 15, 22 of the second cycle and beyond, and rituximab 375 mg/m2 IV was given on day 1, 8, 15, 22 of the second and fifth cycle.

***MYD88* ^{L265P} MUTATION STATUS:** Mutation status was assessed using the previously-described allelespecific polymerase chain reaction, with an analytical sensitivity of approximately 1% mutation detection in a wild-type background.

© 2021 Mayo Foundation for Medical Education and Research

RESULTS

PATIENT POPULATION

- A total of 220 patients with symptomatic previously untreated WM received one of the three regimens as frontline therapy.
- Of these 220 patients, 83 (38%) patients were treated with R-Benda, 92 (42%) patients were treated with DRC while 45 (20%) patients had BDR in the frontline setting.
- One hundred and forty-two (65%) patients were evaluated for *MYD88* ^{L265P} mutation status (R-Benda, n=67; DRC, n=51; BDR, n=24), and of those, 106 (75%) were noted to harbour this mutation; n=59 (88%), n=33 (65%), and n=14 (58%) patients who were treated with R-Benda, DRC or BDR, respectively, p=0.0015.
- The median time from the diagnosis of symptomatic WM to frontline therapy was 0.6 months (95%CI: 0.5-1.0), 0.6 months (95%CI: 0.2-0.7) and 0.4 months (95%CI: 0.1-0.6) for patients who were treated with R-Benda, DRC and BDR, respectively, p=0.74.
- The baseline characteristics of the patients are summarized in **Table 1**.
- Of 220 patients, a small subset of 23 patients received rituximab maintenance therapy following completion of the induction treatment.

TABLE 1: BASELINE CHARACTERISTICS AND LABORATORY PARAMETERS

PARAMETER	R-BENDA	DRC	BDR	P VALUE
Age, median, (range), year	66 (44-87)	65 (36-94)	60 (38-82)	0.14
Sex, Male, n (%)	53 (67)	57 (62)	27 (60)	0.63
Hemoglobin, median (range), g/dL	9.8 (4.6-14.8)	9.5 (5.9-15)	9 (3.6-14)	0.08
Platelets, median, (range), x10^9/dL	196 (30-580)	181 (12-481)	169 (34-460)	0.37
Involved/uninvolved free light-chain ratio	7.2 (0.7-509)	19 (1-973)	42 (2-411)	0.05
IgA, median (interquartile range), mg/dL	51.5 (2-214)	51 (6-781)	52 (5-205)	0.85
IgG, median (interquartile range), mg/dL	609 (44-3290)	589.5 (5-2630)	452 (232-1960)	0.51
IgM, median (interquartile range), mg/dL	3061 (90-10000)	3770 (172-10,500)	4280 (927-63,321)	0.01
M-spike (g/dL)	2.2 (0.2-7.1)	2.5 (0.2-9)	2.9 (0.4-5.9)	0.13
MYD88 ^{L265P} mutation n (%)	59 (88)	33 (65)	14 (58)	**0.001**
IPSS-WM score (n=97)	N=49	N=42	N=6	0.52
Low risk (%)	18	14	0	
Intermediate risk (%)	39	45	67	
High risk (%)	43	40	33	
LDH (U/L)	134.5 (85-338)	154.5 (77-501)	134 (100-180)	0.26
LDH upper limit of normal, >222 U/L, (%)	12.5	20	15	0.19
Beta 2 microglobulin, µg/mL	3.9 (0.8-11.95)	3.2 (1.8-.09)	3.4 (2.4-13.6)	0.37
Marrow LPL cells, median % (range)	72.5 (10-100)	65 (10-95)	60 (10-95)	0.93

R-Benda: rituximab and bendamustine; DRC: dexamethasone, rituximab and cyclophosphamide; BDR: bortezomib, dexamethasone and rituximab; LDH: lactate dehydrogenase; LPL: Lymphoplasmacytic lymphoma

TREATMENT CYCLES AND DOSE MODIFICATIONS

- The median number of cycles was 6 (range: 1-6) for R-Benda, 6 for DRC regimen (range: 1-12) and 6 for BDR (range: 1-12).
- The proportion of patients requiring dose modifications was similar in the three cohorts and was 11%, 7% and 7% in R-Benda, DRC and BDR cohorts, respectively, p=0.58.

TREATMENT EFFICACY AND ENDPOINTS

- The median follow-up for the entire cohort was 4.5 years (95%CI: 4.0-5.0); 2.3 years (95%CI: 2.1-3.2) for the patients treated with R-Benda, 6.3 years (95%CI: 5.5-6.5) for the patients treated with DRC and 5.2 years (95%CI: 5.5-6.5) for patients treated with BDR, p<0.0001.
- Clinically relevant endpoints are depicted in **Figure 1** and **Table 2**.
- Although the proportion of patients receiving rituximab as maintenance was low and similar across the cohorts, in order to assess the impact of induction therapy alone, we performed a subset analysis after excluding the patients who received rituximab maintenance therapy (n=23) following the initial induction therapy and outcomes remained superior for the cohort of patients receiving R-Benda compared to the cohorts that received DRC or BDR regimens.

FIGURE 1

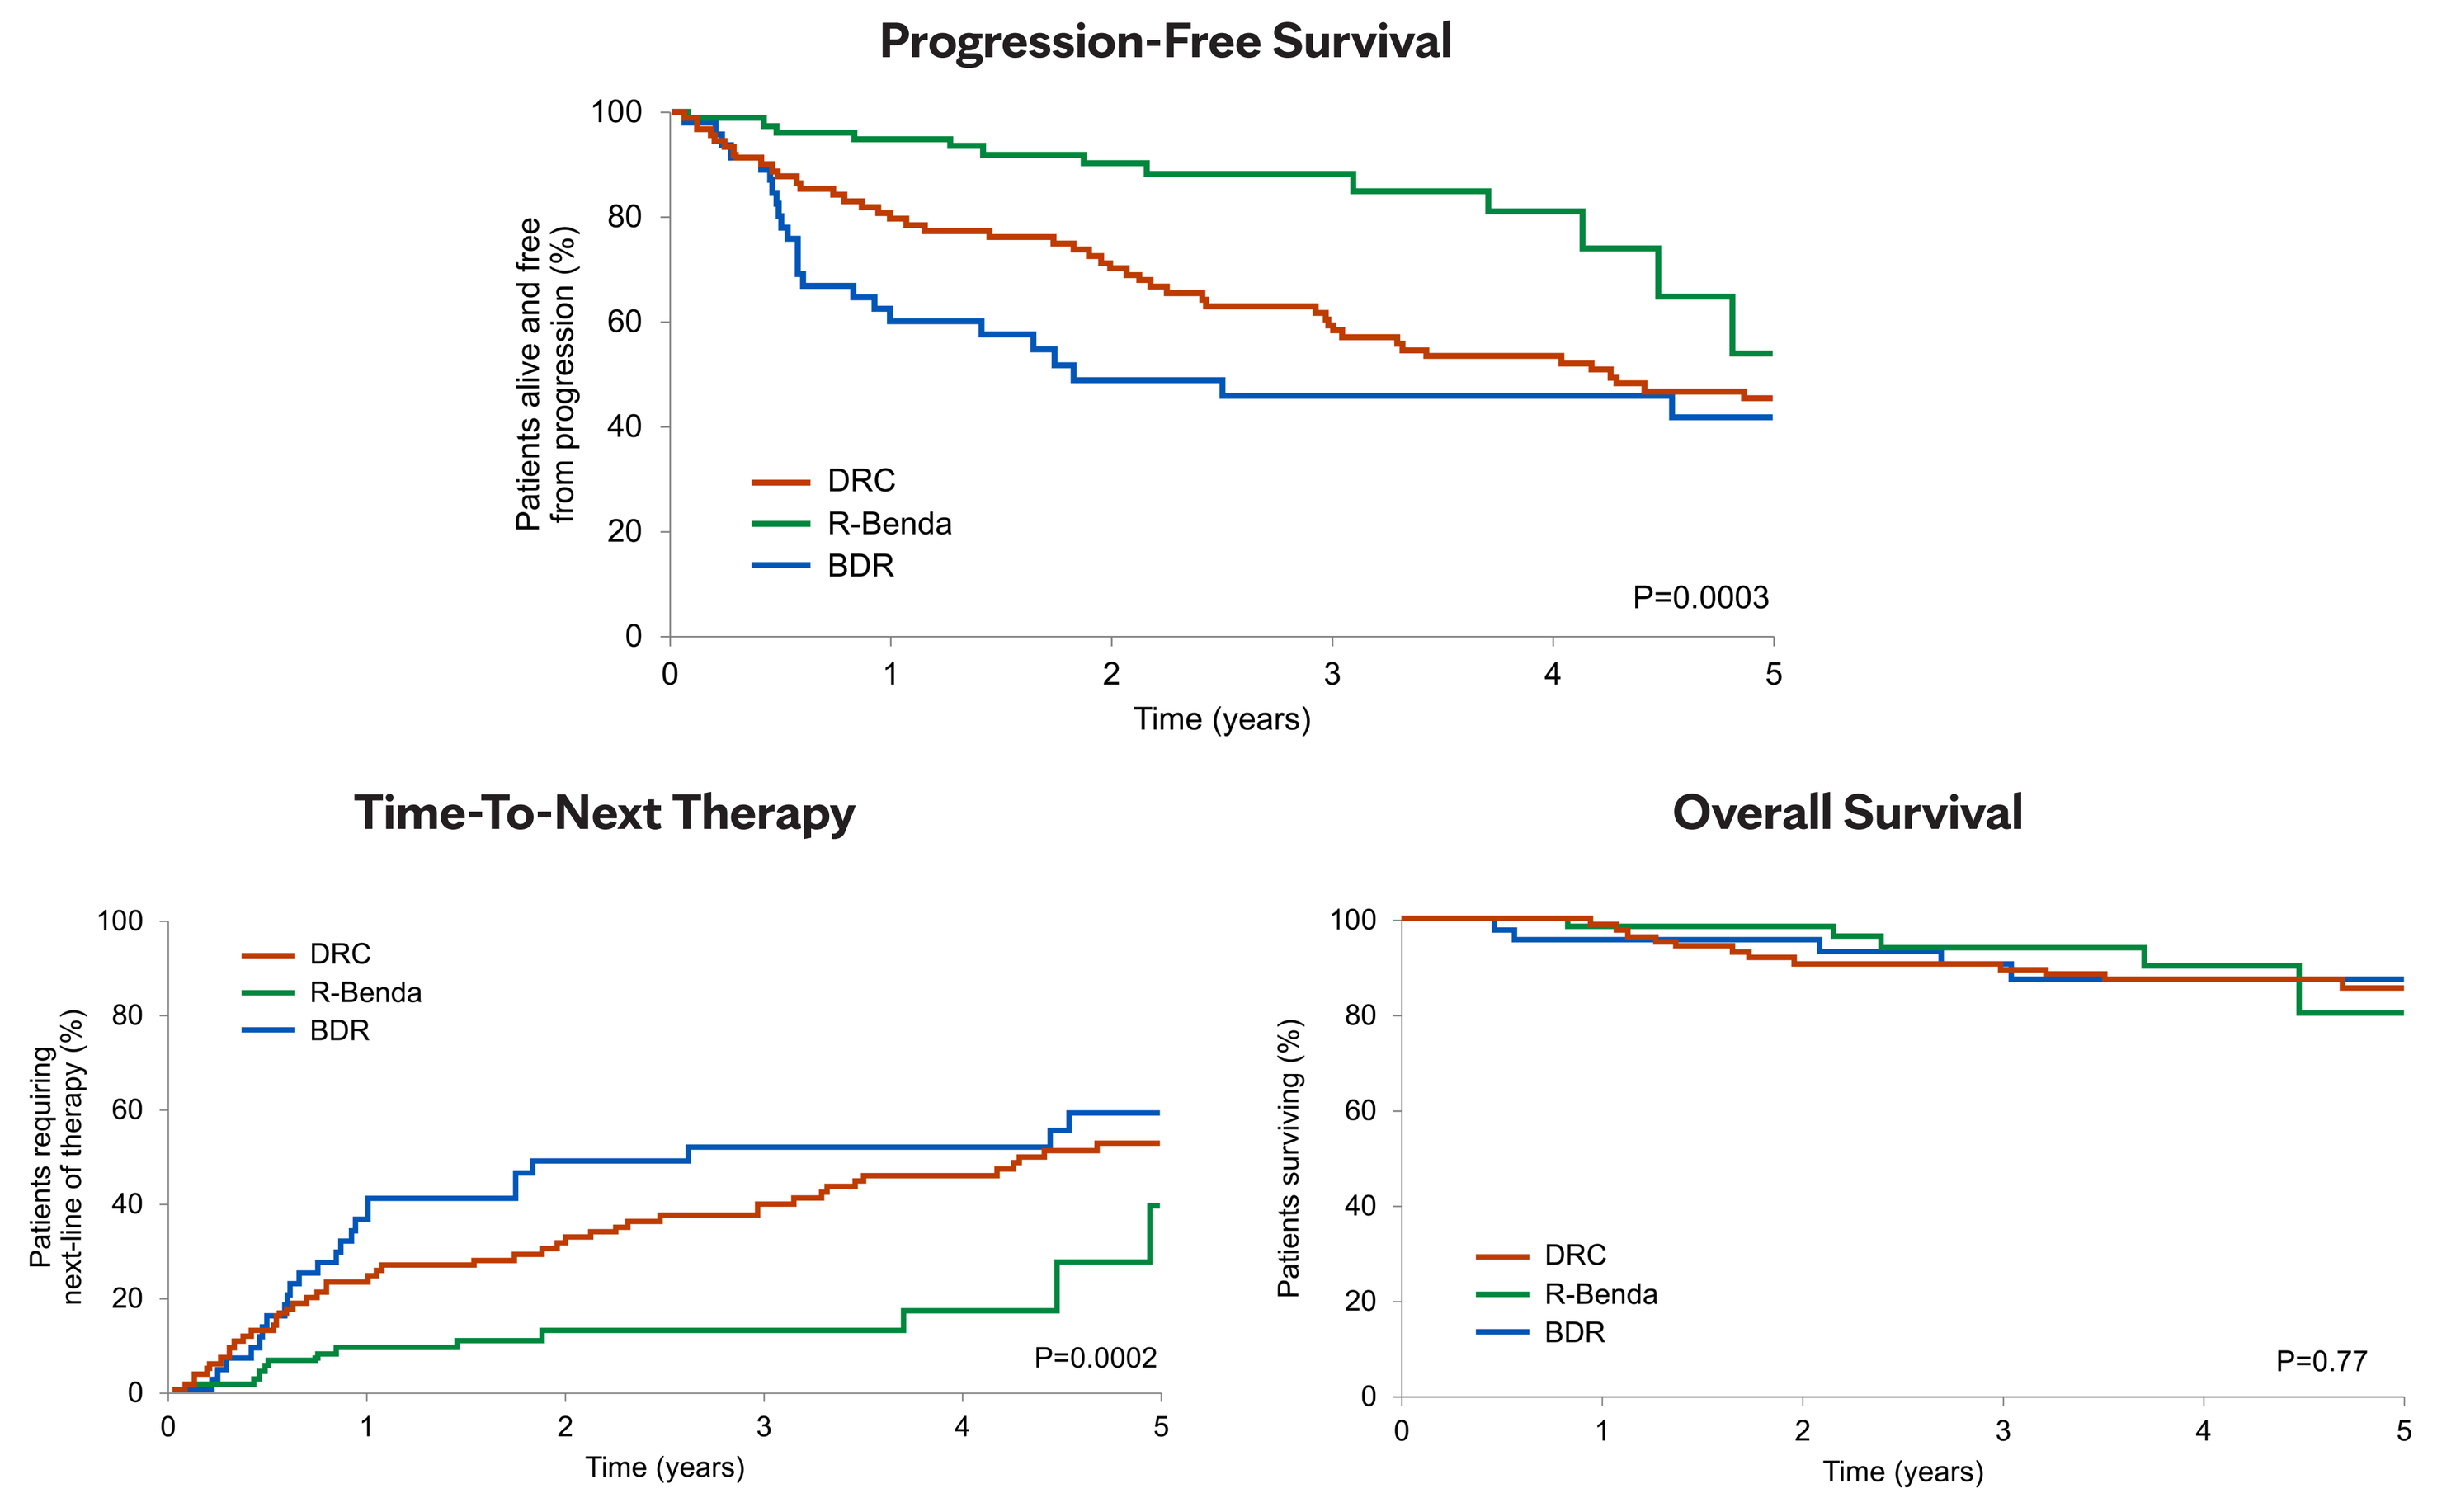
Progression-Free Survival
Patients alive and free from progression (%)
DRC
R-Benda
BDR
P=0.0003
Time (years)
Time-To-Next Therapy
Patients requiring next-line of therapy (%)
DRC
R-Benda
BDR
P=0.0002
Time (years)
Overall Survival
Patients surviving (%)
DRC
R-Benda
BDR
P=0.77
Time (years)

TABLE 2: CLINICALLY RELEVANT ENDPOINTS BASED ON TREATMENT REGIMEN

PARAMETER	R-BENDA	DRC	BDR	P VALUE
Overall response rate (%)	98	78	84	**0.003**
Major response rate (%)	96	53	68	**<0.0001**
Event free survival, range; years (95%CI)	NR (4.4-NR)	4.3 (2.4-5.8)	1.6 (0.6-5.6)	**<0.0001**
Progression free survival, range; years (95%CI)	5.2 (4.5-NR)	4.3 (3.0-5.9)	1.8 (0.8-6.8)	**0.0003**
Time-to-next therapy, range; years (95%CI)	NR (4.5-NR)	4.4 (3.0-5.9)	2.6 (0.9-6.9)	**0.0002**
Time to best response, range; months (95%CI)	4.5 (3.8-5.1)	5.9 (4.8-7.6)	6.7 (5.1-9.0)	**0.005**
Duration of response, range; years (95%CI)	NR (4.5-NR)	3.9 (1.8-6.3)	3.6 (1.0-NR)	**0.001**
Four-year overall survival (%)	90	87	87	0.8

R-Benda: rituximab and bendamustine; DRC: dexamethasone, rituximab and cyclophosphamide; BDR: bortezomib, dexamethasone and rituximab; CI: confidence interval; NR: not-reached. Please refer to supplementary Table 1 for expanded statistical analysis of the treatment related outcomes between the sub-cohorts.

TREATMENT RESPONSE BASED ON *MYD88* ^{L265P} MUTATION STATUS

- Overall, 142 patients were genotyped for *MYD88* mutation.
- Of the 67 patients who received R-Benda and had *MYD88* ^{L265P} genotype data available, the ORR was 98% (n=59) in the *MYD88* ^{L265P} cohort versus 100% (n=8) in the *MYD88* ^{WT} cohort, p=0.6.
- In the DRC cohort with known *MYD88* mutation status, the ORR was 77% (n=33) for patients with *MYD88* ^{L265P} versus 84% (n=18) with *MYD88* ^{WT} genotype, p=0.6.
- Of the 24 patients who received BDR and had undergone *MYD88* ^{L265P} mutational assessment, the ORR was 75% (n=14) in patients with *MYD88* ^{L265P} as compared to 67% (n=10) who had *MYD88* ^{WT} genotype, p=0.7.
- Importantly, the PFS, TTNT, DOR and major response rate (MRR) were not different within treatment groups based on the *MYD88* ^{L265P} mutation status, **Table 3**.

TABLE 3: CLINICALLY RELEVANT ENDPOINTS BASED ON *MYD88* ^{L265P} MUTATION STATUS

PARAMETER	TREATMENT	*MYD88* ^{L265P}	*MYD88* ^{WT}	P VALUE
Overall response rate, (%)	R-Benda DRC BDR	58 (98) 25 (77) 10 (75)	8 (100) 15 (84) 7 (67)	0.6 0.6 0.7
Major response rate, (%)	R-Benda DRC BDR	57 (96) 16 (48) 7 (50)	8 (100) 7 (38) 5 (55)	0.5 0.5 0.8
Time-to-next therapy, range; years (95%CI)	R-Benda DRC BDR	NR (4.9-NR) 3.3 (2.1-4.3) 1.8 (0.6-5.6)	NR (NR-NR) 2.3 (0.5-5.7) 1.4 (0.2-NR)	0.3 0.9 0.8
Time to best response, range; months (95%CI)	R-Benda DRC BDR	4.1 (3.3-5.0) 7.4 (4.2-13.5) 5.7 (3.7-6.7)	5 (2.7-10) 5 (3.7-6.9) 8.5 (2.6-11.8)	0.3 **0.04** 0.06

R-Benda: rituximab and bendamustine; DRC: dexamethasone, rituximab and cyclophosphamide; BDR: bortezomib, dexamethasone and rituximab; CI: confidence interval; NR: not-reached

TREATMENT RELATED TOXICITIES

- The AEs profiles were comparable across the treatment groups, barring neuropathy and neutropenia which were encountered more frequently in the BDR (17.5%) cohort and R-Benda (13%) cohort, respectively.

SALVAGE THERAPY

- Eighty-three (38%) patients received salvage therapy during the follow-up; 10 (12%) in R-Benda cohort, 46 (50%) in DRC cohort and 27 (60%) in the BDR cohort.
- Secondary myeloid neoplasms were not detected in any of the patients on alkylator-rituximab combinations (R-Benda or DRC) so far during the follow-up (median 4.3 years).
- Two patients (2.4%) in the R-Benda cohort went on to require a total of more than three lines of therapy while in the DRC and BDR cohorts, 13 (14%) and 5 (11%), respectively required more than three lines of therapy during their disease course **p=0.01**.

CONCLUSIONS

- The ORR, PFS and TTNT with R-Benda are superior compared to DRC or BDR in treatment-naïve patients with active WM.
- The patient outcomes with any one of these three regimens are unaffected by the *MYD88* ^{L265P} mutation status.