Poster CT-121

Régis Peffault de Latour, MD, PhD; regis.peffaultdelatour@aphp.fr

APPOINT-PNH: Phase 3 Study of the Efficacy and Safety of Iptacopan (LNP023), an Oral Factor B Inhibitor, in Adult Patients With Paroxysmal Nocturnal Hemoglobinuria (PNH) Naive to Complement Inhibitor Therapy (NCT04820530)

Régis Peffault de Latour [1], Bing Han [2], Yasutaka Ueda [3], Georgina Bermann [4], Marion Dahlke [4], Antonio M. Risitano [5,6]

[1]Saint-Louis Hospital, Paris, France; [2]Peking Union Medical College, Beijing, China; [3]Osaka University Graduate School of Medicine, Osaka, Japan; [4]Novartis Pharma AG, Basel, Switzerland; [5]University of Naples Federico II, Naples, Italy; [6]AORN Moscati, Avellino, Italy

Scan to obtain poster

http://novartis.medicalcongressposters.com/Default.aspx?doc=11c97

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

SUMMARY

- There remains an unmet medical need for patients with PNH for a safe and effective treatment inhibiting both intravascular and extravascular hemolysis
- Iptacopan (LNP023) is a first-in-class, oral, small-molecule, selective, and reversible complement factor B inhibitor that has the potential to prevent both intravascular and extravascular hemolysis
- The interim results of 2 ongoing phase 2 studies (NCT03439839, NCT03896152) with iptacopan, both as add-on to eculizumab and as monotherapy, have shown that it is effective and well tolerated at a dose of 200 mg twice daily
- The aim of the phase 3, single-arm, open-label APPOINT-PNH study (NCT04820530) is to determine iptacopan efficacy and safety in treatment-naive patients with PNH who have no history of complement inhibitor therapy, including anti-C5 antibodies

This study is currently recruiting participants, with an estimated primary completion of 2023

This study is sponsored by Novartis Pharmaceuticals Corporation

Presented at the Society of Hematologic Oncology Ninth Annual Meeting; September 8-11, 2021. Virtual.

INTRODUCTION

- Paroxysmal nocturnal hemoglobinuria (PNH) is a rare, acquired hemolytic disorder caused by a mutation in the phosphatidylinositol *N*-acetylglucosaminyltransferase subunit A (*PIGA*) gene and characterized by complement-mediated intravascular hemolysis, bone marrow failure, and severe thrombophilia[1]
- *PIGA*–mutated red blood cells (RBCs) derived from PNH hematopoietic stem cells exhibit a total or partial loss of expression of complement inhibitory proteins CD55 and CD59, resulting in a lack of effective complement inhibition and high susceptibility to complement-mediated hemolysis[1]
- To date, standard of care for PNH has been anti-C5 antibody therapies, but patients continue to experience suboptimal hemoglobin (Hb) responses, extravascular hemolysis, and residual chronic anemia, which can require repeated RBC tranfusions for management and can negatively impact quality of life[2-5]
- Iptacopan (LNP023) is a first-in-class, oral, small-molecule, selective, and reversible complement factor B inhibitor that has the potential to prevent both intravascular and extravascular hemolysis
- The interim results of 2 ongoing phase 2 studies (NCT03439839, NCT03896152) with iptacopan, both as add-on to eculizumab and as monotherapy, have shown that it is efficacious and well tolerated at a dose of 200 mg twice daily[6]
- The purpose of this currently enrolling single-arm, open-label APPOINT-PNH study (NCT04820530) is to determine iptacopan efficacy and safety in treatment-naive patients with PNH who have no history of complement inhibitor therapy, including anti-C5 antibodies

STUDY DESIGN AND METHODS

Key Inclusion and Exclusion Criteria

Figure 1. Key Inclusion and Exclusion Criteria

Key inclusion criteria	Key exclusion criteria
• Age ≥18 years with PNH confirmed by high-sensitivity flow cytometry (RBC and WBC clone size ≥10%)	• Prior treatment with any complement inhibitor, including anti-C5 Abs
• Mean Hb level <10 g/dL	• Active systemic infection (bacterial, viral [including COVID-19], fungal) within 14 days prior to iptacopan administration
• LDH levels >1.5 × ULN	• History of recurrent invasive infections caused by encapsulated organisms, e.g. meningococcus or pneumococcus
• Vaccination against *Neisseria meningitidis* infection is required prior to the start of study treatment	• History of hypersensitivity to iptacopan
	• Known or suspected hereditary complement deficiency
• If not received previously, vaccination against *Streptococcus pneumoniae* and *Haemophilus influenzae* infections should be given	• History of HSC transplant
	• Laboratory evidence of BMF (reticulocytes <100 × 10^9/L; platelets <30 × 10^9/L; neutrophils <0.5 × 10^9/L)

Ab, antibody; BMF, bone marrow failure; Hb, hemoglobin; HSC, hematopoietic stell cell; LDH, lactate dehydrogenase; PNH, paroxysmal nocturnal hemoglobinuria; RBC, red blood cell; ULN, upper limit of normal; WBC, white blood cell.

Study Design

- The study will enroll approximately 40 patients with PNH who have active hemolysis (LDH levels >1.5 × ULN) and anemia (Hb <10 g/dL), and are naive to any complement inhibitor therapy, with approximately 40% of patients having received ≥1 RBC transfusion in the past 6 months
- The study treatment will be iptacopan 200 mg orally twice daily
- After an 8-week screening period, participants will begin a 24-week, single-arm, open-label treatment period for primary efficacy and safety analysis, followed by an optional 24-week, open-label treatment extension period
- After completion of the treatment extension period, participants will be able to join a rollover extension program, which will provide access to iptacopan and enable long-term safety monitoring

Figure 2. Study Design

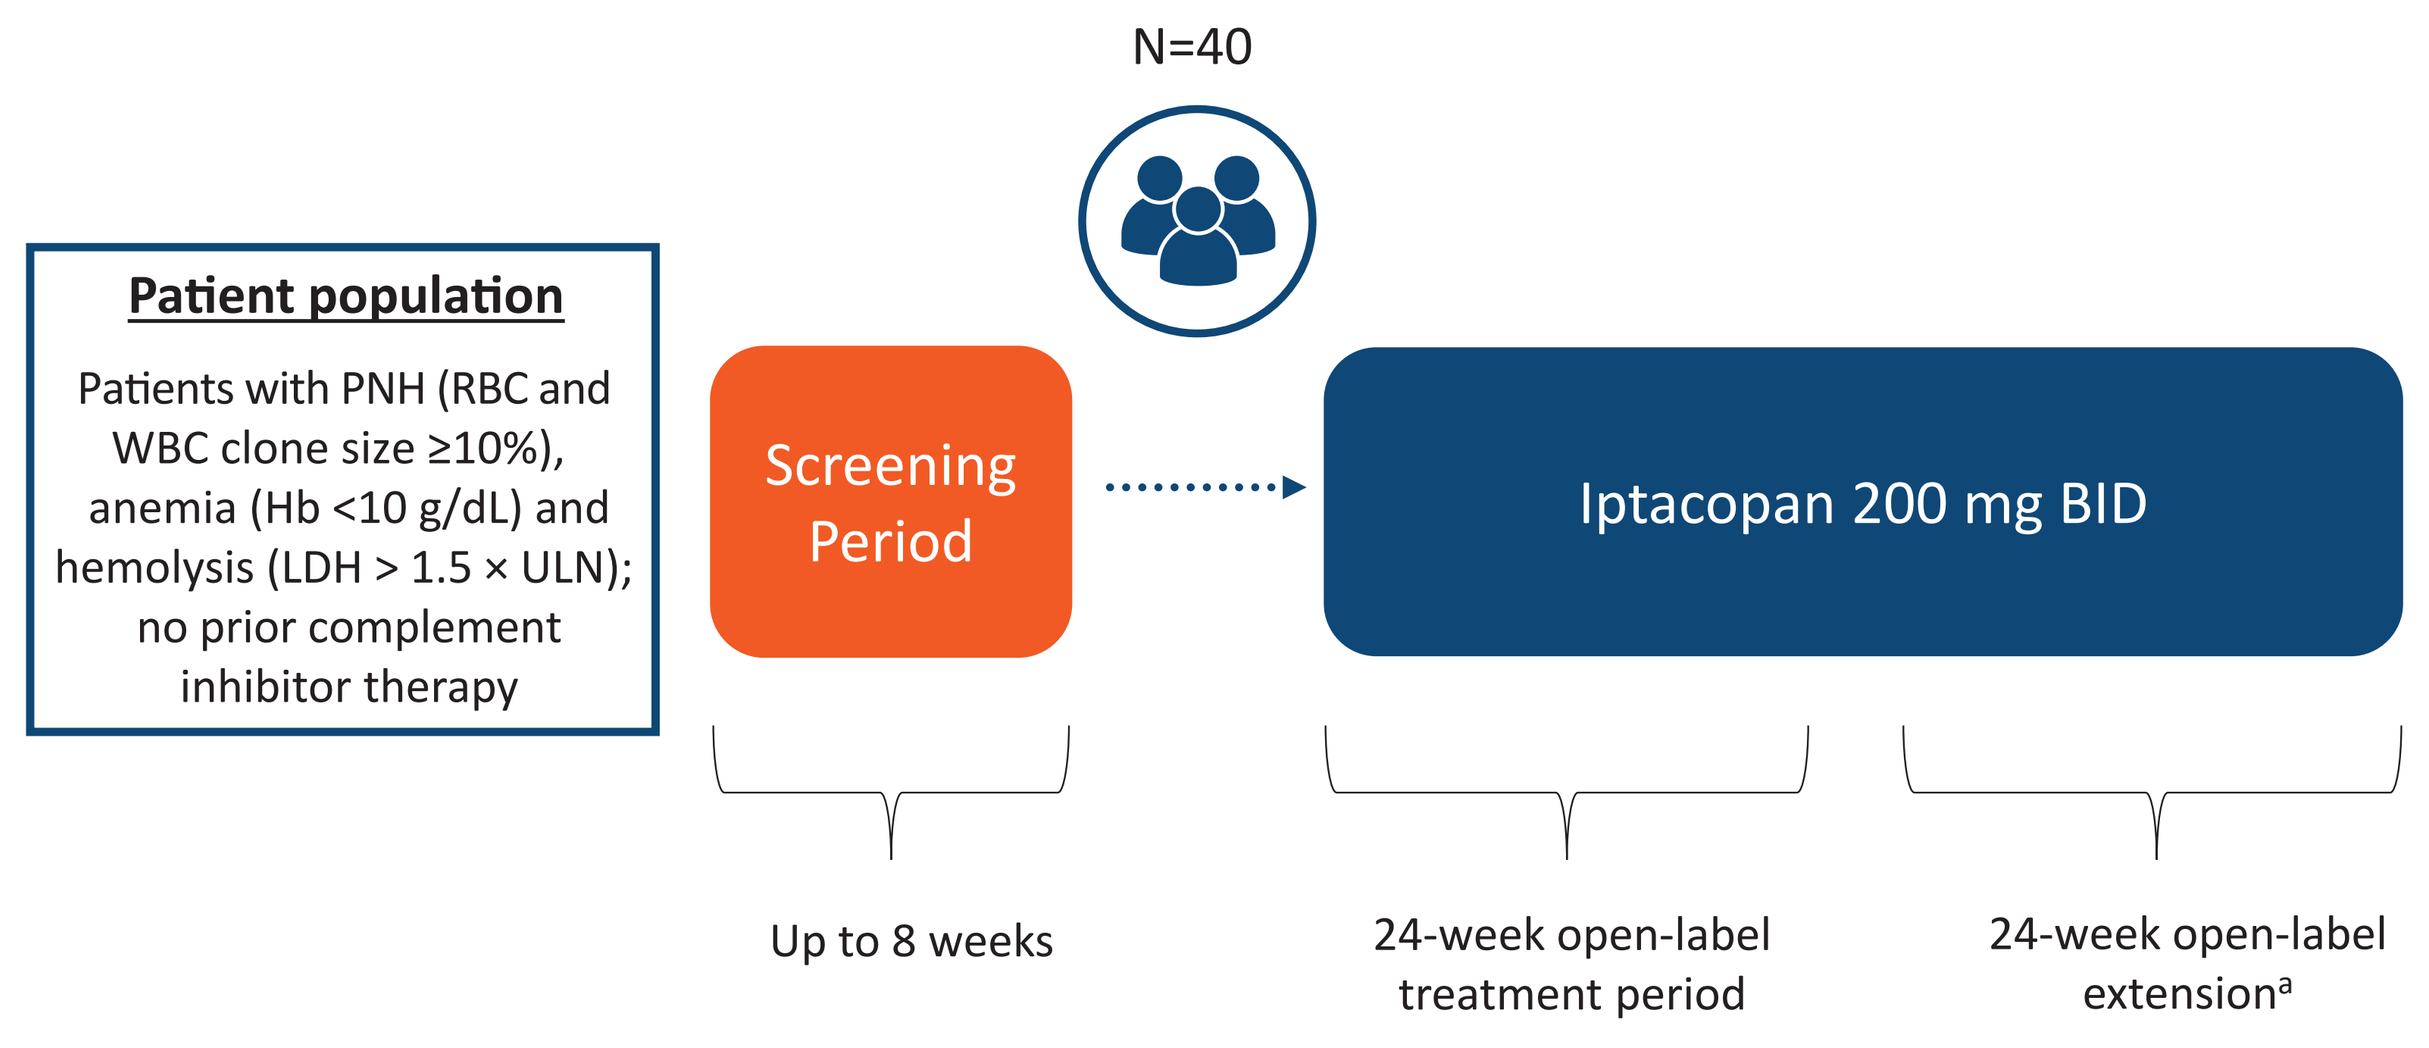
N=40
Patient population
Patients with PNH (RBC and WBC clone size ≥10%), anemia (Hb <10 g/dL) and hemolysis (LDH > 1.5 × ULN); no prior complement inhibitor therapy
Screening Period
Iptacopan 200 mg BID
Up to 8 weeks
24-week open-label treatment period
24-week open-label extension[a]

[a]Patients will have the option to join a roll-over extension program after the 24-week extension period.
BID, twice daily; EOS, end of study; Hb, hemoglobin; LDH, lactate dehydrogenase; PNH, paroxysmal nocturnal hemoglobinuria; RBC, red blood cell; ULN, upper limit of normal; WBC, white blood cell.

Figure 3. APPOINT-PNH Study Sites

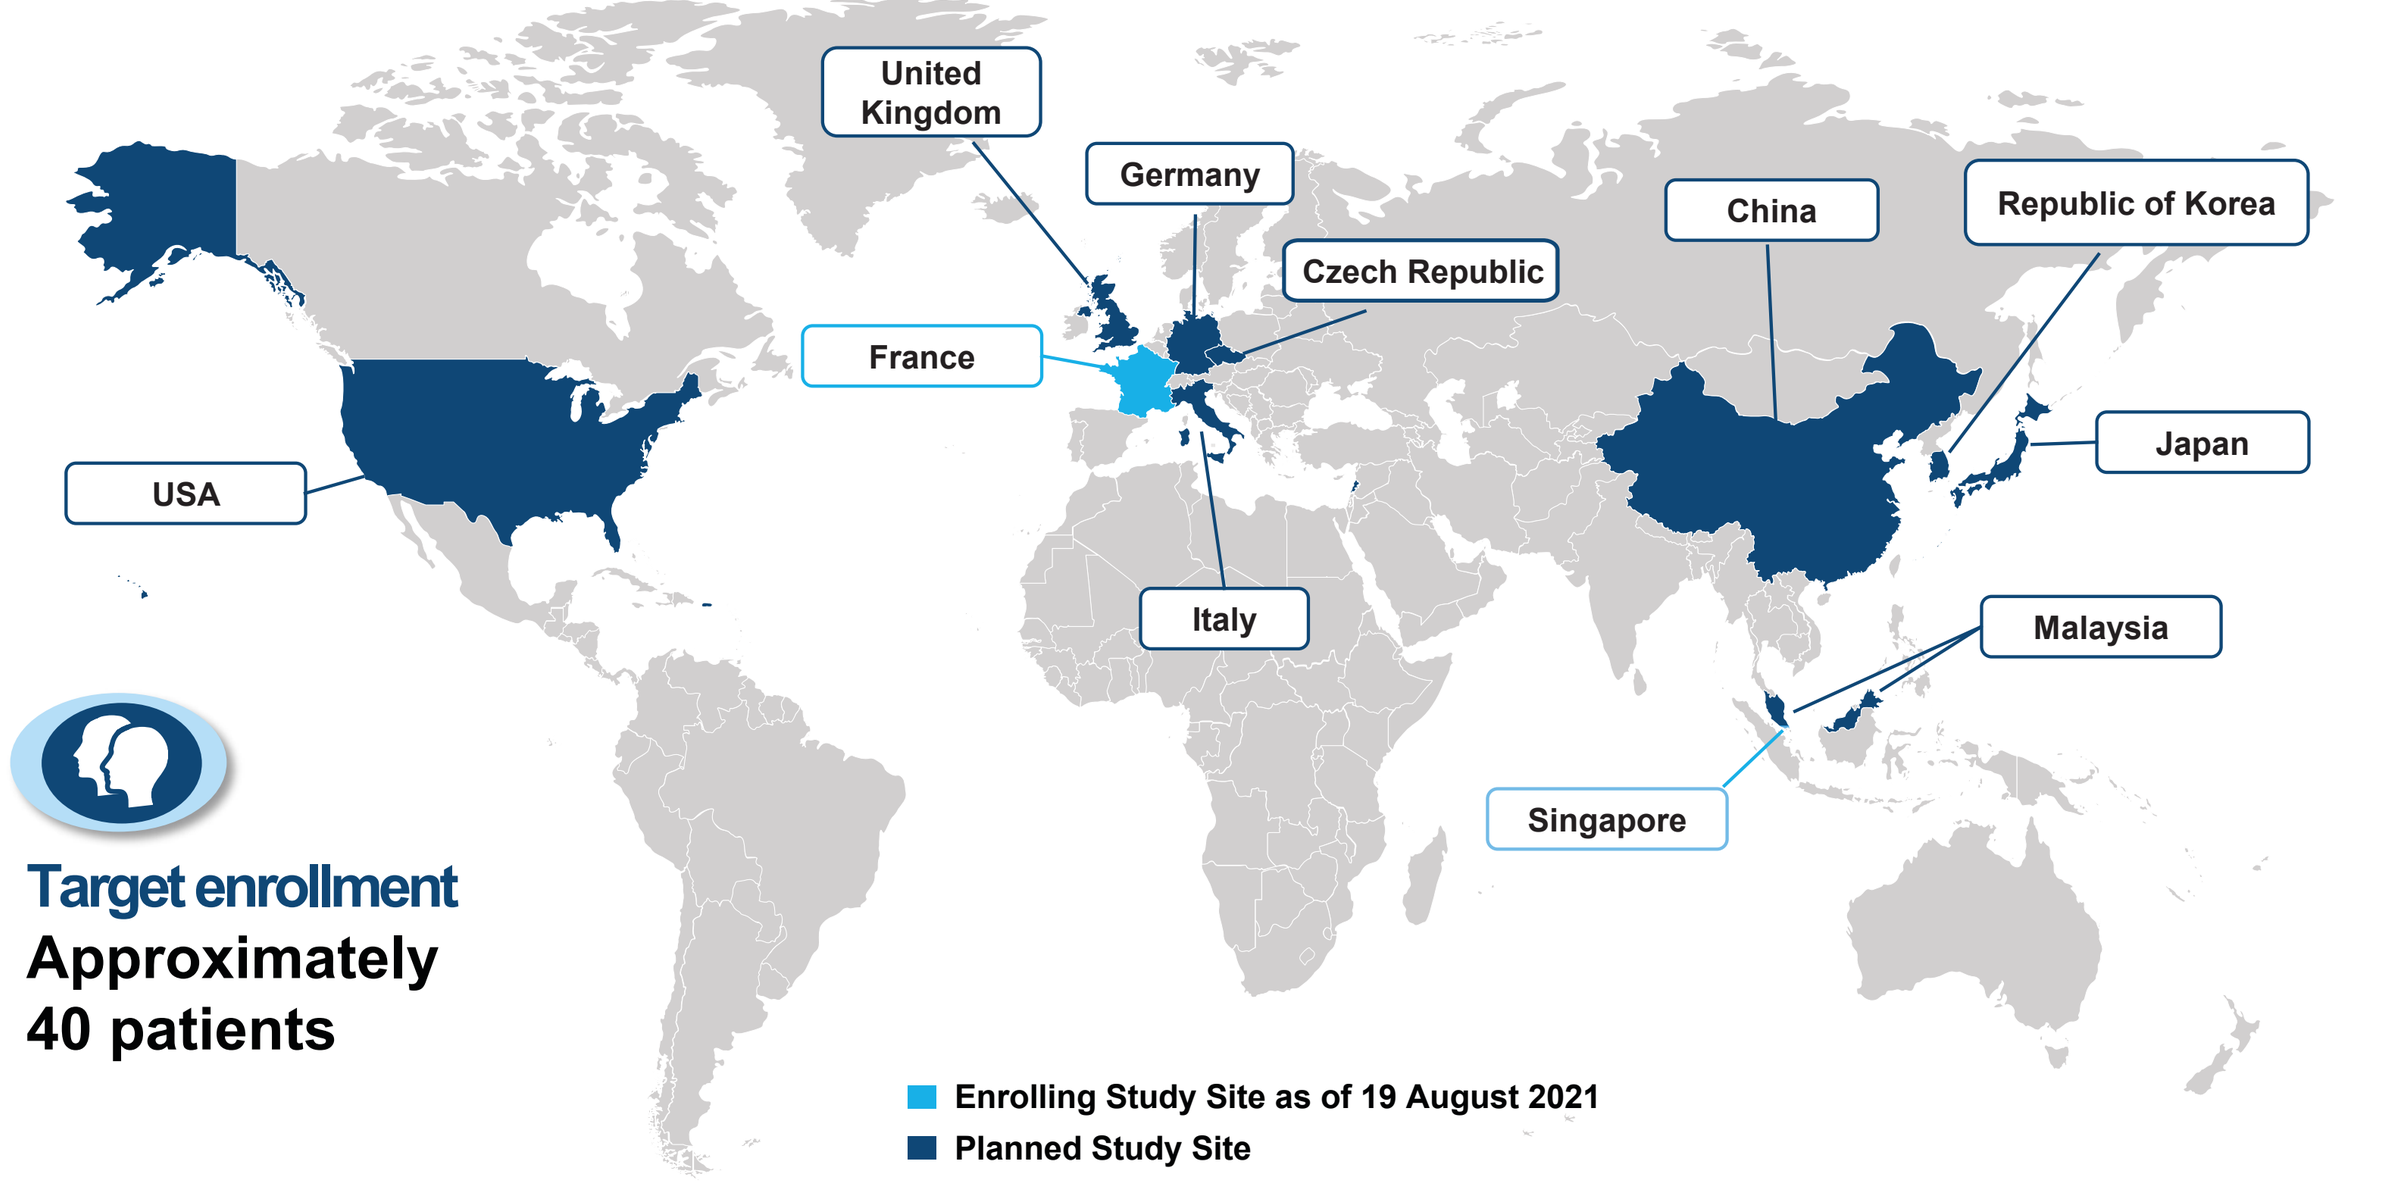
APPOINT-PNH study sites in 11 countries across 3 continents
United Kingdom
Germany
Czech Republic
China
Republic of Korea
France
USA
Japan
Italy
Malaysia
Singapore
Target enrollment
Approximately
40 patients
Enrolling Study Site as of 19 August 2021
Planned Study Site

STUDY OBJECTIVES AND ENDPOINTS

Figure 4. APPOINT-PNH Study Objectives

Primary Objective
- To assess the effect of iptacopan on proportion of patients achieving a sustained increase in Hb levels of ≥2 g/dL from baseline, in the absence of RBC transfusions

Secondary Objectives
- To assess the effect of iptacopan on:
 - Proportion of patients achieving sustained Hb levels ≥12 g/dL, in the absence of RBC transfusions
 - Transfusion avoidance
 - Average change in Hb levels, reticulocyte counts, and percentage change in LDH
 - Improving fatigue, using FACIT-Fatigue
 - Rate of breakthrough hemolysis and MAVEs
- To assess the safety and tolerability of iptacopan

Exploratory Objectives
- To assess the effect of iptacopan on:
 - Hematologic parameters, units of RBC transfusions, and PNH signs and symptoms
 - PRO of overall fatigue severity and health-related QOL
 - PNH clone size and C3d fragment deposition on RBCs
- To characterize the pharmacokinetics of iptacopan
- To assess health care resource utilization
- To explore whether patients experienced meaningful change in fatigue and further feedback on FACIT-Fatigue

Treatment Extension Period Objectives
- To assess the long-term safety, tolerability, and efficacy of iptacopan

FACIT-Fatigue, Functional Assessment of Chronic Illness Therapy-Fatigue Scale; Hb, hemoglobin; LDH, lactate dehydrogenase; MAVE, major adverse vascular event; PNH, paroxysmal nocturnal hemoglobinuria; PRO, patient-reported outcomes; QOL, quality of life; RBC, red blood cell.

Figure 5. APPOINT-PNH Study Endpoints

Primary endpoint
- Response defined as having an increase in Hb levels of ≥2 g/dL from baseline assessed between day 126 and 168 without pRBC transfusions between day 14 and 168

Secondary endpoints
- Response defined as achieving Hb levels ≥12 g/dL between day 126 and 168 in the absence of RBC transfusion between days 14 and 168
- Absence of administration of pRBC transfusions between day 14 and 168
- Change from baseline in Hb (g/dL) as mean of visits between day 126 and 168
- Percent change from baseline in LDH levels as mean of visits between day 126 and 168
- Occurrences of breakthrough hemolysis reported between day 1 and 168
- Change from baseline in reticulocyte counts as mean of visits between day 126 and 168
- Change from baseline in FACIT-Fatigue scores as mean of visits between day 126 and 168
- Occurrences of MAVEs between day 1 and 168
- Safety assessments (including AE/SAEs, safety laboratory parameters, vital signs) between day 1 and 168

Exploratory endpoints
- Hematological parameters, bilirubin levels, units of pRBC transfusions, and PNH signs and symptoms between day 1 and 168
- Change from baseline in patient-reported scores on the EORTC QLQ-C30, EQ-5D-5L, and PGI-S between day 1 and 168
- Percentage of C3d+ RBCs between day 1 and 168
- Type I, Type II & Type III RBCs and PNH clone size (in RBCs and WBCs) between day 1 and 168
- Pharmacokinetic exposure data
- Hospitalizations, readmissions, and ED visits between day 1 and 168
- Patient responses to semi-structured interview focusing on symptoms, particularly fatigue

AE, adverse event; ED, emergency department; EORTC QLQ-C30, European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire Core 30; EQ-5D-5L, EuroQol 5 Dimension 5 Level; FACIT-Fatigue, Functional Assessment of Chronic Illness Therapy - Fatigue Scale; Hb, hemoglobin; LDH, lactate dehydrogenase; MAVE, major adverse vascular events; PGI-S, Patient Global Impression of Severity of Fatigue; PNH, paroxysmal nocturnal hemoglobinuria; pRBC, packed red blood cells; RBC, red blood cell; SAE, serious adverse event; WBC, white blood cell.

ASSESSMENTS

Figure 6: Efficacy and Safety Assessments

Efficacy
- Hb, reticulocytes, LDH, and other PNH-related laboratory parameters
- RBC transfusions
- Breakthrough hemolysis
- PROs (FACIT-Fatigue)
- PNH-related signs and symptoms
- MAVEs

Safety
- Adverse events
- Complete physical examination, vital signs, height and weight, and body temperature
- Laboratory evaluations (full and abbreviated hematologic assessment, full and abbreviated clinical chemistry analysis, urinalysis, UACR, coagulation/markers of thrombosis, pregnancy, blood hormone levels, hepatitis markers, HIV and high-sensitivity flow cytometry [Type I, Type II & Type III erythrocytes, C3d deposition on erythrocytes and PNH clone size in WBCs])
- ECG
- Patients will be given a safety card to monitor for signs and symptoms of infection

Other assessments
- PROs (EORTC QLQ-C30, EQ-5D-5L, and PGI-S)
- Optional patient interviews to understand the extent to which they experienced meaningful changes in PNH-related disease experience and obtain feedback on PROs
- Pharmacokinetics

ECG, electrocardiogram; EORTC QLQ-C30, European Organization for Research and Treatment of Cancer quality of life questionnaire; EQ-5D-5L, EuroQol 5 Dimension 5 Level; FACIT-Fatigue, Functional Assessment of Chronic Illness Therapy-Fatigue Scale; Hb, hemoglobin; LDH, lactate dehydrogenase; MAVE, major adverse vascular event; PGI-S, patient global impression of severity; PNH, paroxysmal nocturnal hemoglobinuria; PRO, patient-reported outcome; RBC, red blood cell; UACR, urine albumin-to-creatinine ratio; WBC, white blood cell.

Statistical Analysis

- The proportion of patients achieving Hb levels ≥2 g/dL in the absence of RBC transfusions, the proportion achieving Hb normalization threshold of ≥12 g/dL, and the proportion achieving transfusion avoidance will be evaluated by means of standardized marginal estimates from a logistic regression model including the following baseline covariates: sex, age (categorical), Hb levels >8 g/dL, and presence of transfusion dependence at enrollment. Confidence intervals will be derived using the bootstrap method
- Mean change from baseline in Hb will impute measurements following transfusions by the low values that would have been observed without transfusions and will be estimated using a repeated-measures model
- Mean changes from baseline FACIT-Fatigue scores and reticulocyte counts, as well as mean percentage change from baseline using log transformed LDH will be derived from a repeated-measures model
- Rates of MAVEs and breakthrough hemolysis will be estimated using a negative binomial model

Acknowledgments

This study is sponsored by Novartis Pharmaceuticals Corporation. We thank Samil Patel, PharmD, from ClinicalThinking, Inc, who provided medical writing support funded by Novartis Pharmaceuticals Corporation in accordance with Good Publication Practice (GPP3) guidelines

Disclosures

RPdL discloses symposium fees & consultancy with Alexion, Novartis, Pfizer and Roche, and research grants from Alexion, Amgen, Novartis and Pfizer; **BH** reports no disclosures to report; **YU** discloses honoraria and consultancy with Alexion, Novartis, Chugai, Sanofi and research funding from Chugai; **GB and MD:** discloses employment with Novartis; **AMR** discloses receiving research support from Alexion Pharmaceuticals, Novartis, Alnylam and Ra Pharma and lecture fees from Alexion, Novartis, Pfizer and Apellis, and served as member of advisory–investigator boards for Alexion, Roche, Achillion, Novartis, Apellis and Samsung, and consulting for Amyndas.

References

1. Brodsky RA. *Blood*. 2014;124(18):2804-2811.
2. DeZern AE, et al. *Eur J Haematol*. 2013;90:16-24.
3. Hill A, et al. *Haematologica*. 2010;95:567-573.
4. McKinley C, et al. *Blood*. 2017;130(suppl 1). Abstract 3471.
5. Risitano AM, et al. *Blood*. 2009;113:4094-4100.
6. Risitano AM, et al. *Lancet Haematol*. 2021;8(5):e344-e354.