Poster CT-118

Phillip Scheinberg, MD; scheinbp@bp.org.br

APPLY-PNH: Phase 3 Study of Efficacy and Safety of Iptacopan (LNP023), an Oral Complement Factor B Inhibitor, in Patients with Paroxysmal Nocturnal Hemoglobinuria and Residual Anemia Despite Anti-C5 Antibody Treatment (NCT04558918)

Antonio M. Risitano [1], Austin Kulasekararaj [2], Carlos M. De Castro [3], Phillip Scheinberg [4], Rafael Levitch [5], Georgina Bermann [5], Marion Dahlke [5], Régis Peffault de Latour [6]

[1]AORN San Giuseppe Moscati, Avellino, Italy; [2]King's College Hospital, National Institute for Health Research/Wellcome King's Clinical Research Facility, and King's College, London, UK; [3]Duke University School of Medicine, Durham, NC; [4]A Beneficência Portuguesa, São Paulo, Brazil; [5]Novartis Pharma AG, Basel, Switzerland; [6]Saint-Louis Hospital, Paris, France

Scan to obtain poster

http://novartis.medicalcongressposters.com/Default.aspx?doc=5fe58

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

SUMMARY

- There remains an unmet medical need for patients with PNH who continue to experience extravascular hemolysis and anemia despite anti-C5 antibody therapy and a need for a safe and effective treatment inhibiting both intravascular and extravascular hemolysis
- Iptacopan (LNP023) is a first-in-class, oral, small-molecule, selective, and reversible complement factor B inhibitor that has the potential to prevent both intravascular and extravascular hemolysis
- The interim results of 2 ongoing phase 2 studies (NCT03439839, NCT03896152) with iptacopan, both as add-on to eculizumab and as monotherapy, have shown that it is effective and well tolerated at a dose of 200 mg twice daily
- The aim of the phase 3, randomized, multicenter, active comparator-controlled, open-label APPLY-PNH trial (NCT04558918) is to demonstrate the superiority of iptacopan in patients with residual anema despite anti-C5 antibody therapy using novel efficacy endpoints
- This study will be the first head-to-head comparison of iptacopan vs an anti-C5 monoclonal antibody in patients with PNH

This study is currently recruiting participants, with an estimated primary completion of 2022

This study is sponsored by Novartis Pharmaceuticals Corporation

Presented at the Society of Hematologic Oncology Ninth Annual Meeting; September 8-11, 2021. Virtual.

INTRODUCTION

- Paroxysmal nocturnal hemoglobinuria (PNH) is a rare, acquired hemolytic disorder caused by a mutation in the phosphatidylinositol *N*-acetylglucosaminyltransferase subunit A (*PIGA*) gene and characterized by complement-mediated intravascular hemolysis, bone marrow failure, and severe thrombophilia[1]
- *PIGA*–mutated red blood cells (RBCs) derived from PNH hematopoietic stem cells exhibit a total or partial loss of expression of complement inhibitory proteins CD55 and CD59, resulting in a lack of effective complement inhibition and high susceptibility to complement-mediated hemolysis[1]
- Thus far, anti-C5 antibody treatment has been generally effective in treating intravascular hemolysis; however, some patients do not to achieve hemoglobin (Hb) normalization and they experience C3-mediated extravascular hemolysis, leading to symptomatic PNH despite anti-C5 antibody treatment[2,3]
- Iptacopan (LNP023) is a first-in-class, oral, small-molecule, selective, and reversible complement factor B inhibitor that has the potential to prevent both intravascular and extravascular hemolysis
- The interim results of 2 ongoing phase 2 studies (NCT03439839, NCT03896152) with iptacopan, both as add-on to eculizumab and as monotherapy, have shown that it is effective and well tolerated at a dose of 200 mg twice daily[4]
- The purpose of the phase 3, randomized, multicenter, active comparator-controlled, open-label APPLY-PNH trial (NCT04558918) is to determine whether iptacopan is superior to anti-C5 antibody therapy in the treatment of patients with PNH presenting with residual anemia

STUDY DESIGN AND METHODS

Key Inclusion and Exclusion Criteria

Figure 1. Key Inclusion and Exclusion Criteria

Key inclusion criteria	Key exclusion criteria
Age ≥18 years with PNH confirmed by high-sensitivity flow cytometry (RBC and WBC clone size ≥10%)	Treatment with eculizumab with dosing interval ≤11 days
Stable anti-C5 Ab treatment for ≥6 months before randomization	Known or suspected hereditary complement deficiency
Mean Hb level <10 g/dL	History of hypersensitivity to the study drugs
Vaccination against *Neisseria meningitidis* infection is required prior to the start of treatment	History of HSC transplant
If not received previously, vaccination against *Streptococcus pneumoniae* and *Haemophilus influenzae* infections should be given	Laboratory evidence of BMF (reticulocytes $<100 \times 10^9$/L; platelets $<30 \times 10^9$/L; neutrophils $<500 \times 10^6$/L)
	Active systemic bacterial, viral (incl. COVID-19), or fungal infection within 14 days prior to iptacopan administration
	History of recurrent infections caused by encapsulated organisms (e.g. meningococcus or pneumococcus)

Ab, antibody; BMF, bone marrow failure; Hb, hemoglobin; HSC, hematopoietic stem cell; PNH, paroxysmal nocturnal hemoglobinuria; RBC, red blood cell; WBC, white blood cell.

Study Design

- The study will enroll approximately 91 patients with PNH with residual anemia, defined as Hb <10 g/dL despite stable regimen of anti-C5 antibody treatment in the last 6 months prior to randomization, with approximately 40% of patients having received at least 1 packed RBC transfusion in the past 6 months
- After an 8-week screening period, patients will be randomized 8:5 to receive iptacopan 200 mg twice daily or anti-C5 antibody therapy (patients continuing with the same regimen during the randomized treatment period as they were on prior to randomization), stratified by prior anti-C5 antibody treatment (eculizumab or ravulizumab) and transfusion history during the 6 months prior to randomization
- After 24 weeks, patients in the anti-C5 antibody arm may switch to iptacopan for a 24-week extension period if they wish to participate, and those receiving iptacopan may also continue their treatment for the extension period
- After completion of the treatment extension period, the participants will be able to join the rollover extension program, which will provide access to iptacopan and enable long-term safety monitoring

Figure 2. Study Design

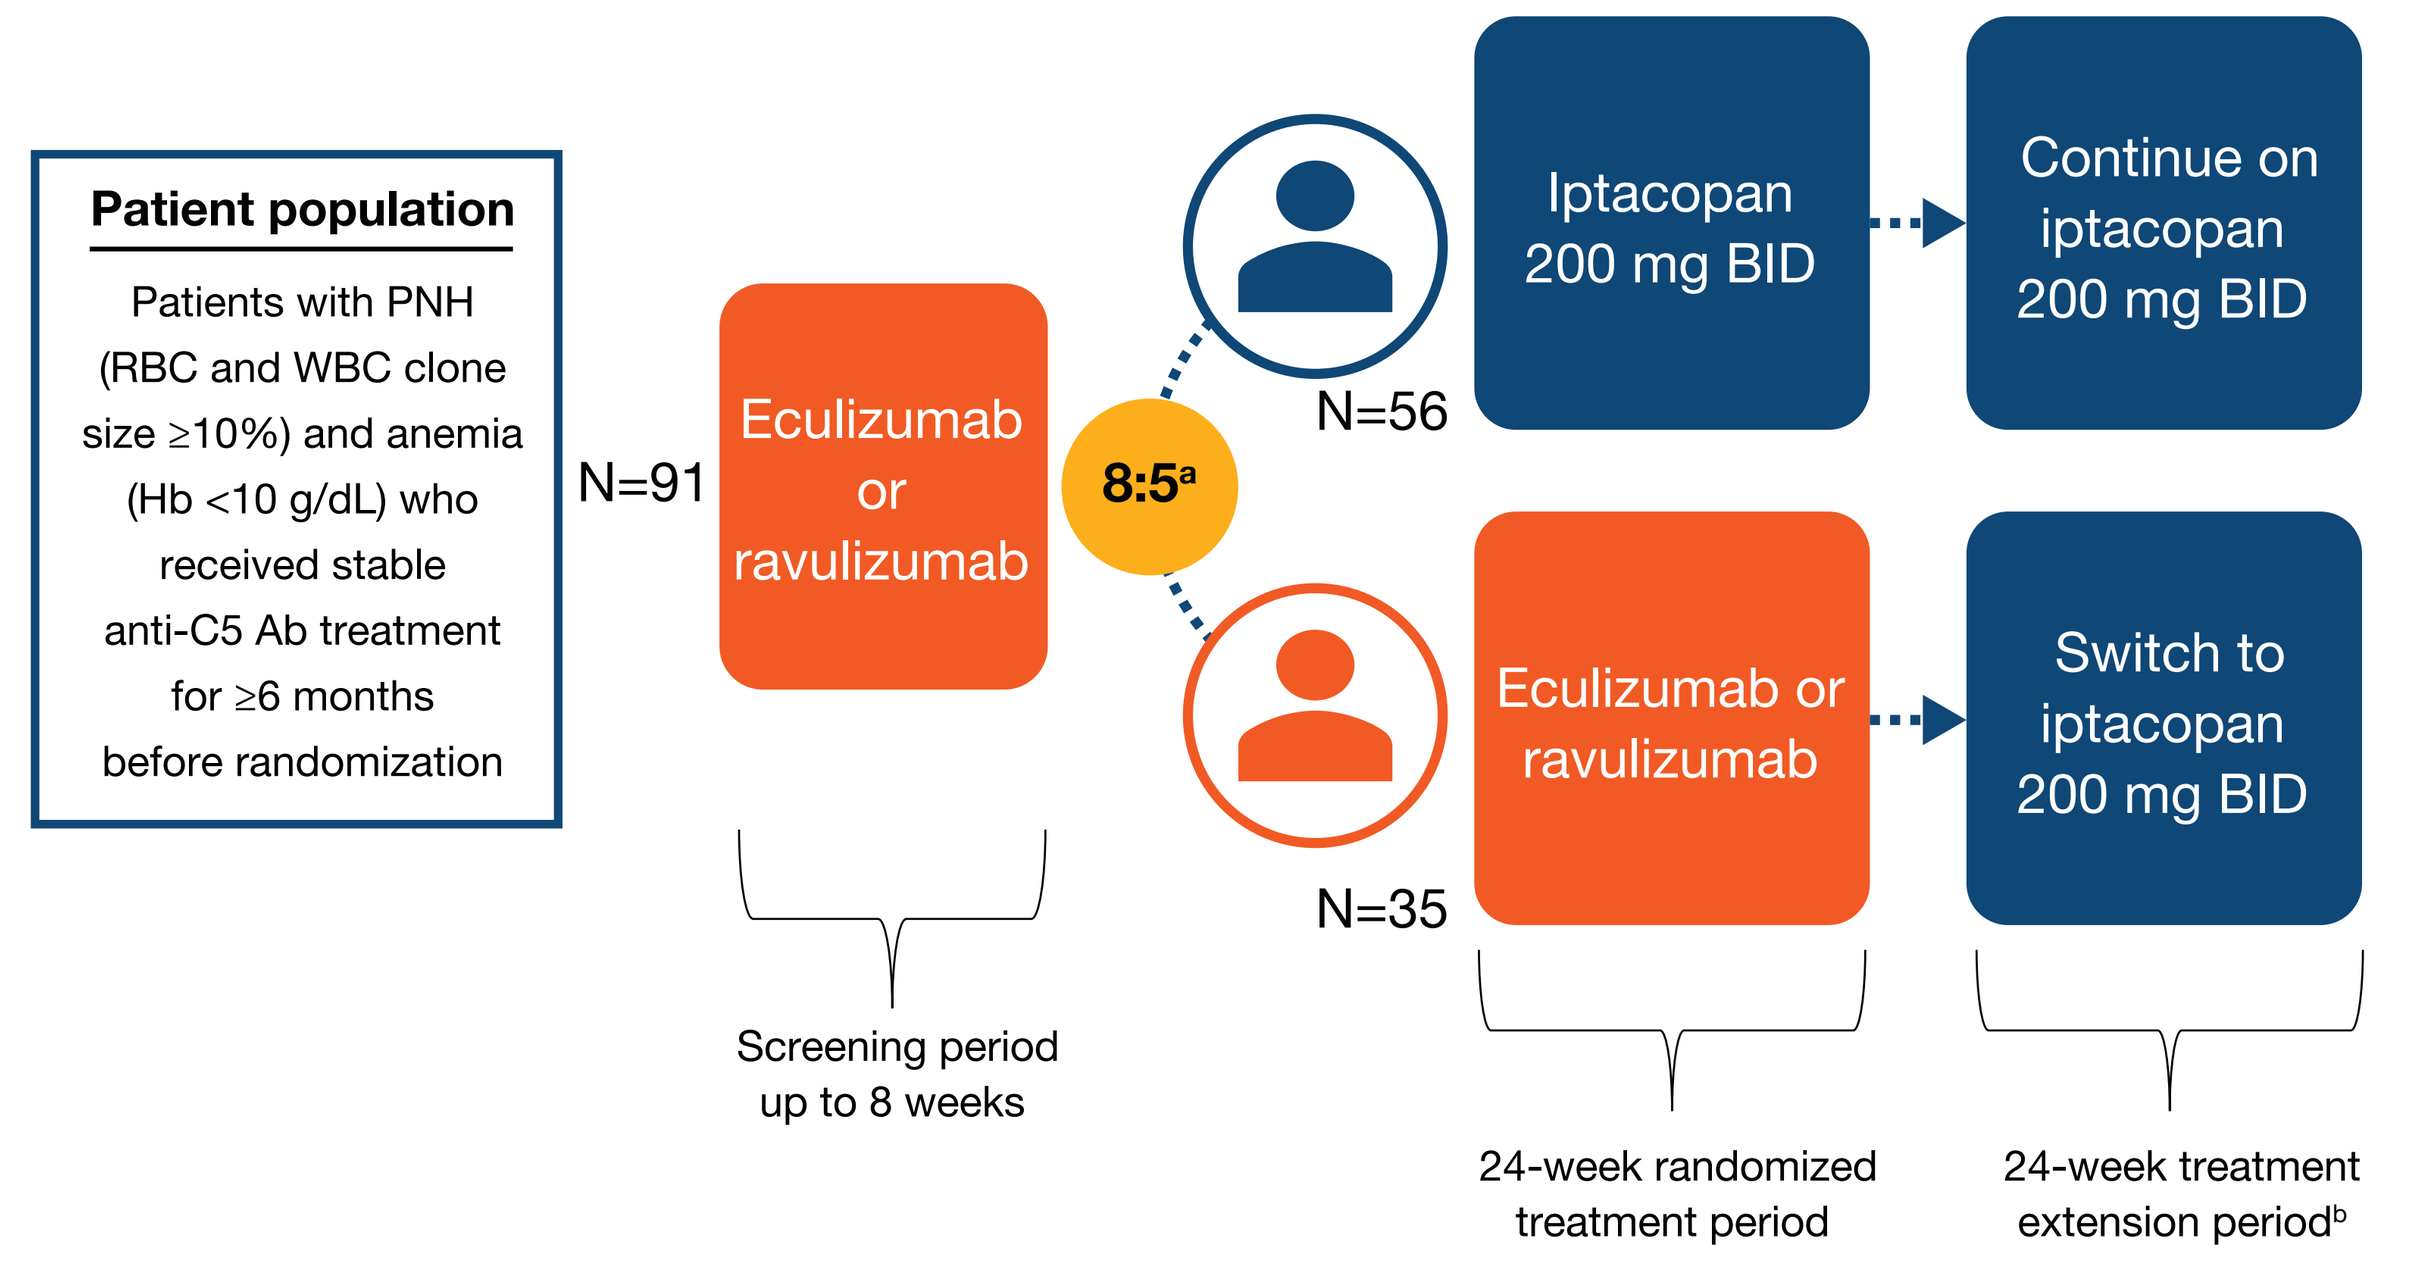
Patient population
Patients with PNH (RBC and WBC clone size ≥10%) and anemia (Hb <10 g/dL) who received stable anti-C5 Ab treatment for ≥6 months before randomization
N=91
Eculizumab or ravulizumab
8:5[a]
N=56
Iptacopan 200 mg BID
Continue on iptacopan 200 mg BID
N=35
Eculizumab or ravulizumab
Switch to iptacopan 200 mg BID
Screening period up to 8 weeks
24-week randomized treatment period
24-week treatment extension period[b]

[a]There should be a seamless switch with an overlap of at least 1 (eculizumab) or 2 (ravulizumab) weeks of prior anti-C5 Ab and iptacopan treatment.
[b]Patients will have the option to join a rollover extension program after the 24-week extension period.
Ab, antibody; BID, twice daily; Hb, hemoglobin; PNH, paroxysmal nocturnal hemoglobinura; RBC, red blood cell; WBC, white blood cell.

Figure 3. APPLY-PNH Study Sites

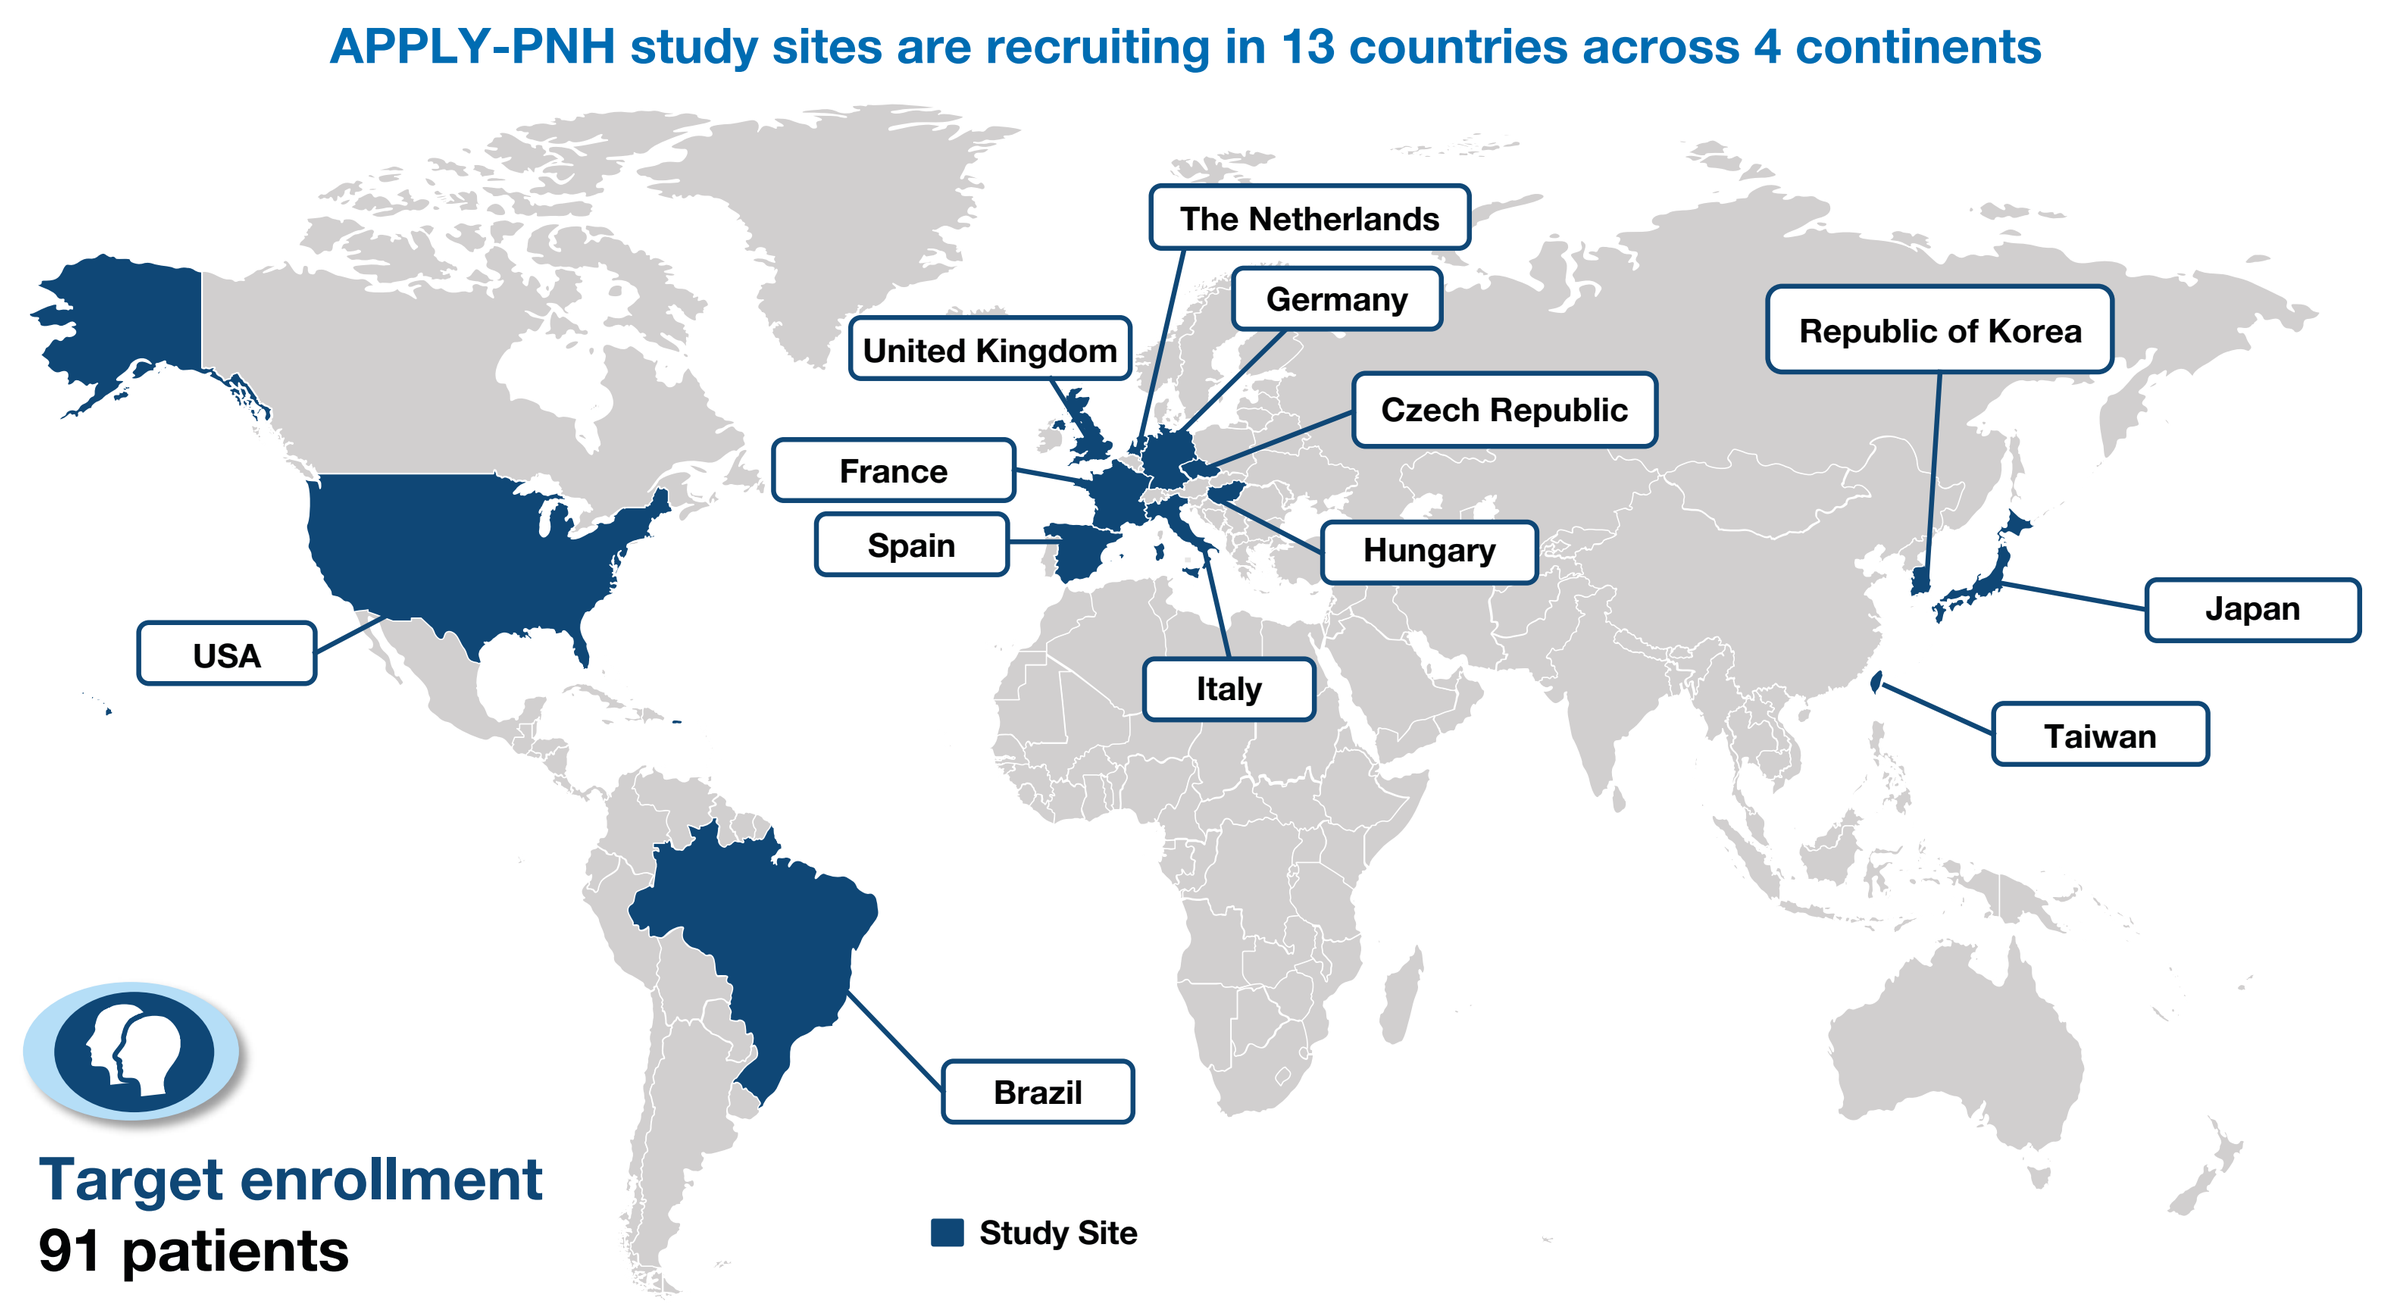
APPLY-PNH study sites are recruiting in 13 countries across 4 continents
The Netherlands
Germany
United Kingdom
Republic of Korea
Czech Republic
France
Spain
Hungary
USA
Japan
Italy
Taiwan
Brazil
Target enrollment
91 patients
Study Site

STUDY OBJECTIVES AND ENDPOINTS

Figure 4. APPLY-PNH Study Objectives

Primary Objectives
- To demonstrate superiority of iptacopan compared with anti-C5 Ab treatment in the proportion of participants achieving a sustained increase in Hb levels of ≥2 g/dL from baseline in the absence of RBC transfusions
- To demonstrate superiority of iptacopan vs anti-C5 Ab treatment in the proportion of participants achieving sustained Hb levels of ≥12 g/dL in the absence of RBC transfusions

Secondary Objectives
- To demonstrate the superiority of iptacopan compared with anti-C5 Ab treatment in:
 - Transfusion avoidance
 - Average change in Hb levels
 - Improving fatigue
 - Average change in reticulocyte counts
 - Percentage change in LDH
 - Rate of breakthrough hemolysis
- To assess the rates of MAVEs of iptacopan compared with anti-C5 Ab treatment
- To assess the safety and tolerability of iptacopan compared with anti-C5 Ab treatment

Exploratory Objectives
- To assess the effect of iptacopan compared with anti-C5 Ab treatment in:
 - Hematologic parameters units of RBC transfusions, and PNH signs and symptoms
 - PRO of overall fatigue severity and health-related QOL
 - C3d fragment deposition on RBCs and PNH clone size
 - Utilization of health care resources
- To characterize the pharmacokinetics of iptacopan in patients with PNH
- To explore whether patients experienced meaningful change in fatigue and further feedback on FACIT-Fatigue

Treatment Extension Period Objective
- To assess the long-term safety, tolerability, and efficacy of iptacopan

Ab, antibody; FACIT-Fatigue, Functional Assessment of Chronic Illness Therapy – Fatigue Scale; Hb, hemoglobin; LDH, lactate dehydrogenase; MAVEs, major adverse vascular events; PNH, paroxysmal nocturnal hemoglobinura; PRO, patient reported outcomes; QOL, quality of life; RBC, red blood cell.

Figure 5: APPLY-PNH Endpoints

Primary endpoints
- Response defined as having an increase in Hb levels of ≥2 g/dL from baseline assessed between day 126 and 168 in the absence of pRBC transfusions between day 14 and 168
- Response defined as having Hb ≥12 g/dL between day 126 and 168 in the absence of packed RBC transfusions between day 14 and 168

Secondary endpoints
- Absence of administration of pRBC transfusions between day 14 and 168
- Change from baseline in Hb (g/dL) as a mean of visits between day 126 and 168
- Change from baseline in FACIT-Fatigue scores as a mean of visits between day 126 and 168
- Change from baseline in reticulocyte counts as a mean of visits between day 126 and 168
- Percentage change from baseline in LDH levels as a mean of visits between day 126 and 168
- Occurrence of breakthrough hemolysis between day 1 and 168
- Occurrence of MAVEs between day 1 and 168
- Safety assessments (including AE/SAEs, safety laboratory parameters, vital signs)

Exploratory endpoints
- Hematological parameters, bilirubin levels, units of pRBC transfusions, and PNH signs and symptoms between day 1 and 168
- Change from baseline in patient-reported scores on the EORTC QLQ-C30, EQ-5D-5L, and PGI-S between day 1 and 168
- Percentage of C3d+ RBCs between day 1 and 168
- Type I, Type II & Type III RBCs and PNH clone size (in RBCs and WBCs) between day 1 and 168
- Pharmacokinetic exposure data
- Hospitalizations, readmissions, and ED visits between day 1 and 168
- Patient responses to semi-structured interview focusing on symptoms, particularly fatigue

AE, adverse event; ED, emergency department; EORTC QLQ-C30, European Organisation for Research and Treatment of Cancer Quality of Life Questionnaire Core 30; EQ-5D-5L, EuroQol 5 Dimension 5 Level; FACIT-Fatigue, Functional Assessment of Chronic Illness Therapy - Fatigue Scale; Hb, hemoglobin; LDH, lactate dehydrogenase; MAVE, major adverse vascular events; PGI-S, Patient Global Impression of Severity of Fatigue; PNH, paroxysmal nocturnal hemoglobinuria; pRBC, packed red blood cells; RBC, red blood cell; SAE, serious adverse event; WBC, white blood cell.

ASSESSMENTS

Figure 6: Efficacy and Safety Assessments

Efficacy
- Hb, reticulocytes, LDH, and other PNH-related laboratory parameters
- RBC transfusions
- Breakthrough hemolysis
- PROs (FACIT-Fatigue)
- PNH-related signs and symptoms
- MAVEs

Safety
- Adverse events
- Complete physical examination, vital signs, height and weight, and body temperature
- Laboratory evaluations (full and abbreviated hematologic assessment, full and abbreviated clinical chemistry analysis, urinalysis, UACR, coagulation/markers of thrombosis, pregnancy, blood hormone levels, hepatitis markers, HIV, and high-sensitivity flow cytometry [Type I, Type II & Type III erythrocytes, C3d deposition on erythrocytes and PNH clone size in WBCs])
- ECG
- Patients will be given a safety card to monitor for signs and symptoms of infection

Other assessments
- PROs (EORTC QLQ-C30, EQ-5D-5L, and PGI-S)
- Optional patient interviews to understand the extent to which they experienced meaningful changes in PNH-related disease experience and obtain feedback on PROs
- Pharmacokinetics
- Biomarkers

ECG, electrocardiogram; EORTC QLQ-C30, European Organisation for Research and Treatment of Cancer Quality of Life questionnaire core 30; EQ-5D-5L, EuroQol 5 Dimension 5 Level; FACIT-Fatigue, Functional Assessment of Chronic Illness Therapy-Fatigue Scale; Hb, hemoglobin; LDH, lactate dehydrogenase; MAVE, major adverse vascular event; PGI-S, Patient Global Impression of Severity; PNH, paroxysmal nocturnal hemoglobinuria; PRO, patient-reported outcome; RBC, red blood cell; UACR, urine albumin to creatinine ratio; WBC, white blood cell.

Statistical Analysis

- Potential superiority of iptacopan vs anti-C5 antibody in the analysis of the primary endpoints will be tested by an odds ratio from a conditional logistic regression model, accounting for stratification factors used at randomization and adjusted for age, sex, and baseline Hb level
- The study α will be split equally between the 2 primary endpoints and will determine rejection of each based on the permutation distribution of the *P*-values
- The α from the test of primary endpoints is passed down to the secondary endpoints with more weight given to the significance level of transfusion avoidance followed by change from baseline in Hb, change in total score of FACIT-Fatigue, and change from baseline in reticulocyte counts, recycling α between these after rejecting each hypothesis
- If these are successfully tested in favor of iptacopan, percent change from baseline in LDH, occurrence of breakthrough hemolysis, and occurrence of MAVEs (including thrombosis) will be tested at full study α

Acknowledgments

This study is sponsored by Novartis Pharmaceuticals Corporation. We thank Michael Cikovic, PharmD, from ClinicalThinking, Inc, who provided medical writing support funded by Novartis Pharmaceuticals Corporation in accordance with Good Publication Practice (GPP3) guidelines

Disclosures

AMR discloses receiving research support from Alexion Pharmaceuticals, Novartis, Alnylam and Ra Pharma and lecture fees from Alexion, Novartis, Pfizer and Apellis, and served as member of advisory–investigator boards for Alexion, Roche, Achillion, Novartis, Apellis and Samsung, and consulting for Amyndas. **AK** discloses receiving funding from Alexion for speaking at educational meetings. **CMDC** discloses serving as a consultant to Alexion. **PS** discloses consulting AbbVie, Alexion Pharmaceuticals, Janssen, Celgene, and Novartis. **RL, GB, and MD** disclose employment with Novartis Pharma AG. **RPdL** discloses symposium fees & consultancy with Alexion, Novartis, Pfizer and Roche, and research grants from Alexion, Amgen, Novartis and Pfizer.

References

1. Brodsky RA. *Blood*. 2014;124(18):2804-2811.
2. Risitano AM, et al. *Blood*. 2009;113(17):4094-4100.
3. McKinley C, et al. *Blood*. 2017;130(suppl 1). Abstract 3471.
4. Risitano AM, et al. *Lancet Haematol*. 2021;8(5):e344-e354.