CML-413

Jorge Cortes, Jorge.cortes@augusta.edu

Asciminib Provides Durable Responses and a Favorable Safety Profile in Patients With Chronic Myeloid Leukemia (CML) in Chronic Phase (CP) With the T315I Mutation in a Phase I Study

Jorge E. Cortes,[1] Timothy P. Hughes,[2] Michael J. Mauro,[3] Andreas Hochhaus,[4] Delphine Réa,[5] Yeow-Tee Goh,[6] J.J.W.M. Janssen,[7] Michael C. Heinrich,[8] Moshe Talpaz,[9] Gabriel Etienne,[10] Massimo Breccia,[11] Michael Deininger,[12] Philipp le Coutre,[13] Fabian Lang,[14] Fotis Polydoros,[15] Silvia Cacciatore,[15] Laura Stenson,[16] Dong-Wook Kim[17]

[1]Georgia Cancer Center, Augusta, GA; [2]South Australian Health and Medical Research Institute and University of Adelaide, Adelaide, Australia; [3]Memorial Sloan Kettering Cancer Center, New York, NY; [4]Universitätsklinikum Jena, Jena, Germany; [5]Hôpital Saint-Louis, Paris, France; [6]Singapore General Hospital, Bukit Merah, Singapore; [7]VU University Medical Center, Amsterdam, the Netherlands; [8]Veterans Affairs Portland Health Care System and Oregon Health and Science University Knight Cancer Institute, Portland, OR; [9]University of Michigan Comprehensive Cancer Center, Ann Arbor, MI; [10]The University of Bordeaux, Bordeaux, France; [11]Università la Sapienza, Rome, Italy; [12]Huntsman Cancer Institute, University of Utah, Salt Lake City, UT; [13]Charité - Universitätsmedizin Berlin, Berlin, Germany; [14]Department of Medicine, Hematology and Oncology, Goethe University Hospital, Frankfurt, Germany; [15]Novartis Pharma AG, Basel, Switzerland; [16]Novartis Ireland Limited, Dublin, Ireland; [17]Seoul St. Mary's Hospital, College of Medicine, Catholic University of Korea, Seoul, South Korea

Scan to obtain:
- Poster
- Supplementary Podcast

http://novartis.medicalcongressposters.com/Default.aspx?doc=948e9
Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

KEY FINDINGS & CONCLUSIONS

- Asciminib 200 mg twice daily (BID) has a favorable safety profile and meaningful clinical efficacy in patients with the T315I mutation
- Nearly half of the patients achieved major molecular response (MMR), which has been durable in most of the patients
- MMR was achieved by ponatinib-naive and -pretreated patients, with a higher incidence in ponatinib-naive patients
- The safety profile of asciminib 200 mg BID is consistent with that observed at a lower dose in patients without the T315I mutation
 - Lipase level increase and thrombocytopenia were the most common grade 3/4 adverse events (AEs)
 - Arterial occlusive AEs were reported in 3 patients
- Asciminib is a promising therapeutic option for patients with CML-CP/accelerated phase (AP) with the T315I mutation, including those for whom ponatinib treatment has failed

This study is sponsored by Novartis Pharma AG.
Poster presented at: SOHO Annual Meeting; September 8-11, 2021; Houston, TX.

INTRODUCTION

- The *BCR-ABL1* T315I mutation confers resistance to all approved adenosine triphosphate (ATP)–competitive tyrosine kinase inhibitors (TKIs) except ponatinib; compound mutations involving T315I can also confer resistance to ponatinib[1-3]
- Asciminib has a different mechanism of action from available TKIs. Asciminib has been designated as the 1st STAMP (**S**pecifically **T**argeting the **A**BL1 **M**yristoyl **P**ocket) inhibitor (**Figure 1**)[3-6]
- In earlier results from this study, asciminib showed clinical activity and a favorable safety profile in patients with the T315I mutation[6,7]

Figure 1: BCR-ABL1 Structure and TKI Binding Sites

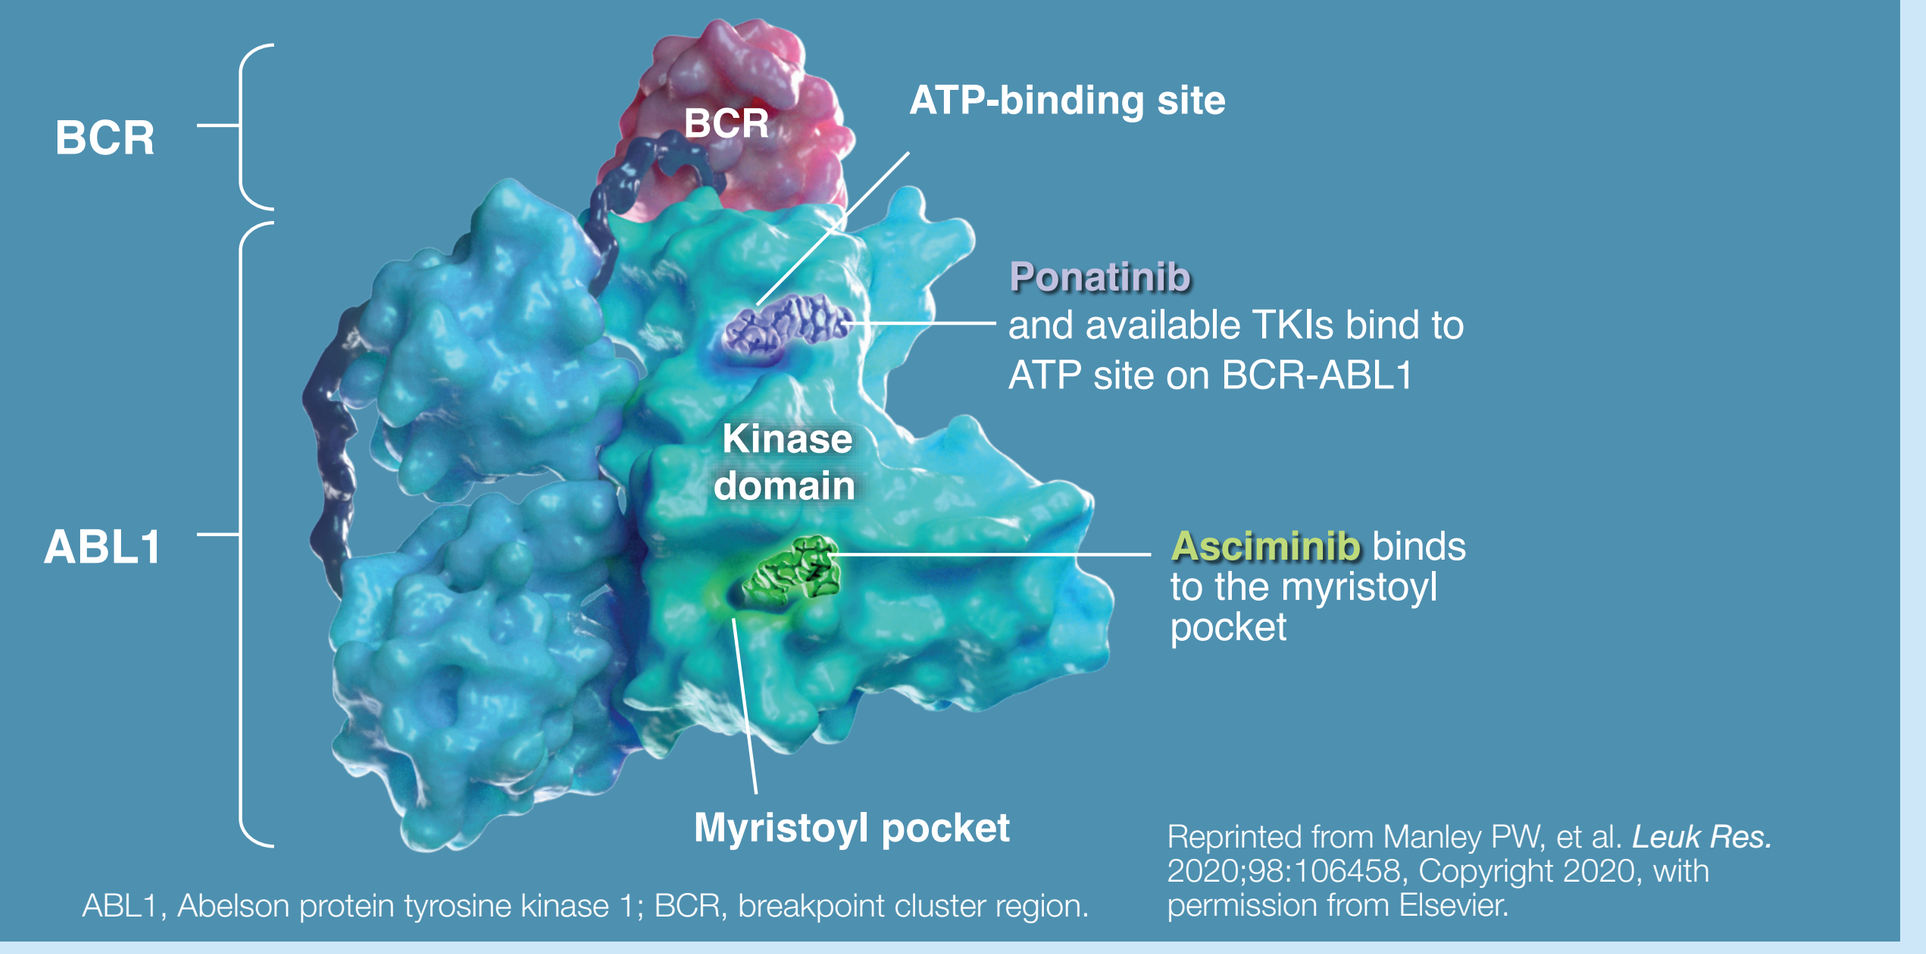
BCR
ABL1
BCR
ATP-binding site
Ponatinib and available TKIs bind to ATP site on BCR-ABL1
Kinase domain
Asciminib binds to the myristoyl pocket
Myristoyl pocket

ABL1, Abelson protein tyrosine kinase 1; BCR, breakpoint cluster region.

Reprinted from Manley PW, et al. *Leuk Res.* 2020;98:106458, Copyright 2020, with permission from Elsevier.

METHODS

- This is a phase I, multicenter, open-label, dose-finding study of asciminib in patients with CML or Philadelphia chromosome (Ph)–positive acute lymphoblastic leukemia (ALL) who have been treated with ≥2 prior TKIs (**Figure 2**)
- The primary objective of this study was to determine the maximum tolerated dose or recommended dose
- Eligible patients for this study were aged ≥18 years with CML-CP/AP or Ph+ ALL
- Here we report updated efficacy and safety results from the expansion cohort in patients with CML-CP/AP and the T315I mutation treated with asciminib 200 mg BID

Figure 2. Study Design

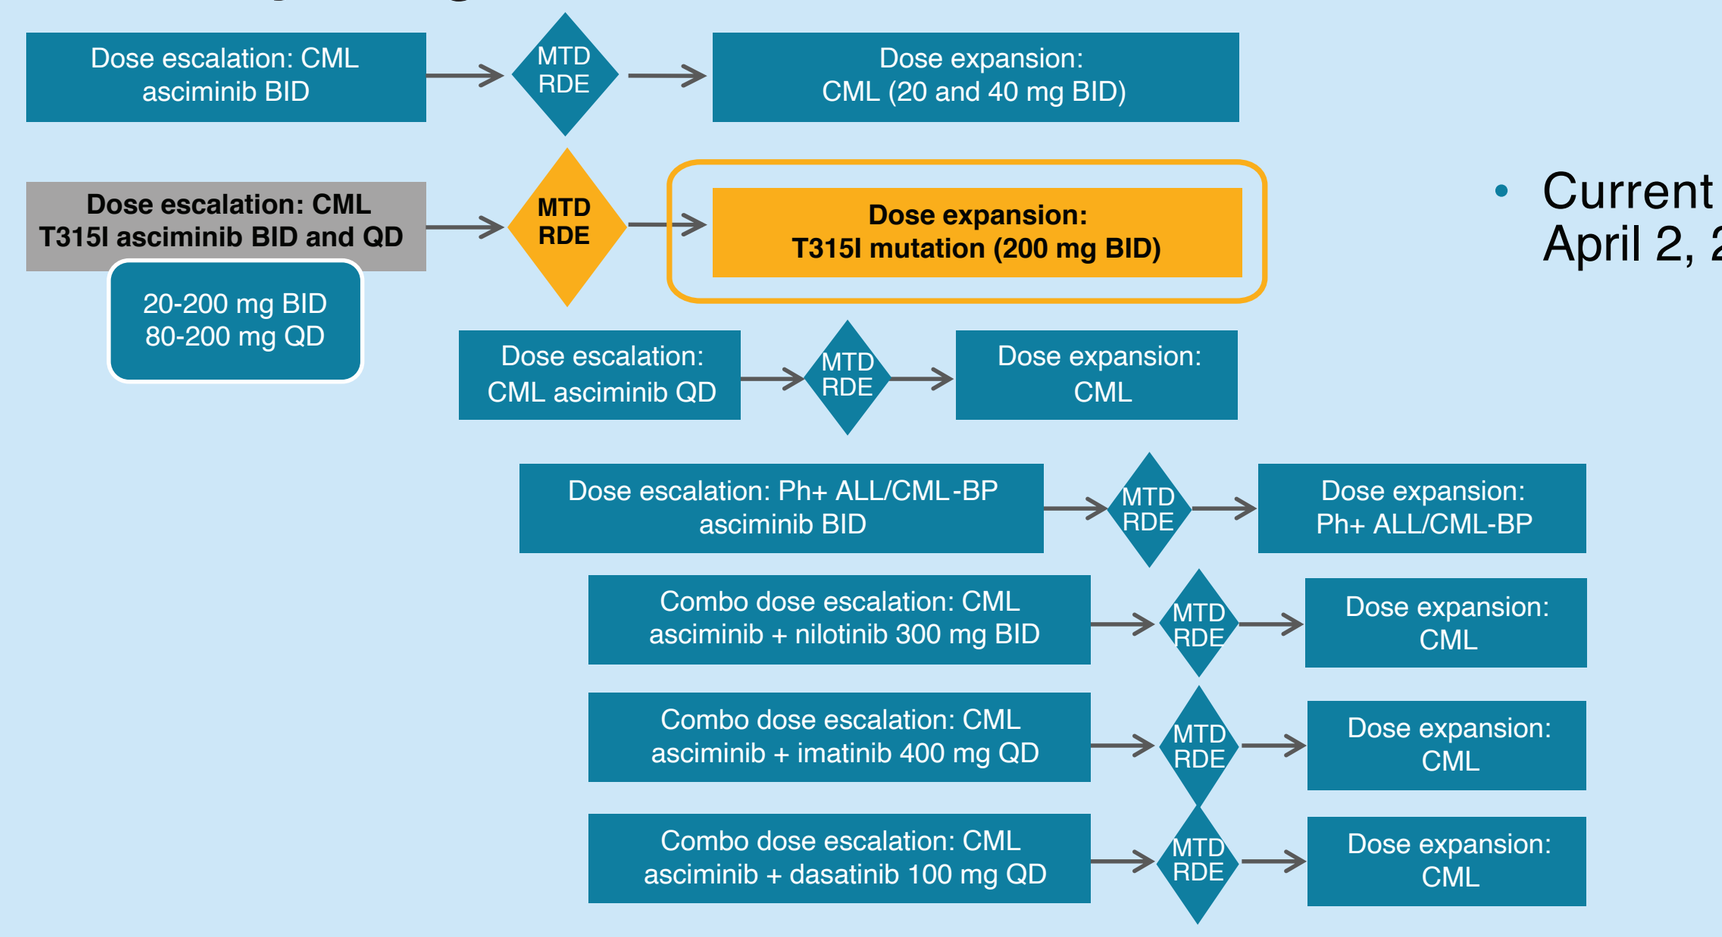
Dose escalation: CML asciminib BID
MTD RDE
Dose expansion: CML (20 and 40 mg BID)
Dose escalation: CML T315I asciminib BID and QD
20-200 mg BID 80-200 mg QD
MTD RDE
Dose expansion: T315I mutation (200 mg BID)
Dose escalation: CML asciminib QD
MTD RDE
Dose expansion: CML
Dose escalation: Ph+ ALL/CML-BP asciminib BID
MTD RDE
Dose expansion: Ph+ ALL/CML-BP
Combo dose escalation: CML asciminib + nilotinib 300 mg BID
MTD RDE
Dose expansion: CML
Combo dose escalation: CML asciminib + imatinib 400 mg QD
MTD RDE
Dose expansion: CML
Combo dose escalation: CML asciminib + dasatinib 100 mg QD
MTD RDE
Dose expansion: CML

- Current analysis is based on a data cutoff of April 2, 2020, and is focused on the cohort in gold

From Hughes TP, et al. *N Engl J Med*. 2019;381(24):2315-2326. Copyright © 2019 Massachusetts Medical Society. Reprinted with permission from Massachusetts Medical Society.

BP, blast phase; combo, combination; MTD, maximum tolerated dose; QD, once daily; RDE, recommended dose for expansion.

RESULTS

Table 1. Baseline Characteristics

Characteristic	N=52
Age, median (range), years	54 (26-86)
Male, n (%)	39 (75.0)
ECOG performance status, n (%)	
0	39 (75.0)
1	13 (25.0)
CML phase, n (%)	
CP	48 (92.3)
No. of prior TKIs, n (%)	
1	9 (17.3)
2	16 (30.8)
3	18 (34.6)
≥4	9 (17.3)
Prior TKIs, n (%)	
Dasatinib	35 (67.3)
Ponatinib	31 (59.6)
Nilotinib	30 (57.7)
Imatinib	29 (55.8)
Bosutinib	4 (7.7)
Time from diagnosis, median (range), years	3.9 (0.5-27.9)
Mutations at screening [VAF], n (%)[a]	
T315I alone	49 (94.2)
T315I and E255K [90/20]	1 (1.9)
T315I and E355G [100/100]	1 (1.9)
T315I and F359I [80/30]	1 (1.9)
BCR-ABL1^{IS} at screening, n (%)[b]	
>0.1% to ≤1%	8 (15.4)
>1% to ≤10%	13 (25.0)
>10%	28 (53.8)
Atypical/unknown transcripts[c]	3 (5.8)

ECOG, Eastern Cooperative Oncology Group; IS, International Scale; VAF, variant allele frequency.
[a] All mutations detected using Sanger sequencing or next-generation sequencing methods with an allele frequency >0 are reported.
[b] Overall, 49 patients were evaluable for molecular response analysis because 3 patients had atypical/unknown transcripts at screening.
[c] These were ponatinib-pretreated patients with e13a3 transcripts, e19a2 transcripts, and unknown transcript type (n=1 each).

- Ponatinib was the most recent prior TKI in 28 of 31 ponatinib-pretreated patients patients (**Table 1**)
- Ponatinib was discontinued due to intolerance (n=10), resistance (n=15), and other reasons (n=6)
- Ponatinib-pretreated patients were more heavily pretreated than ponatinib-naive patients (**Table 2**)

Table 2. Characteristics of Ponatinib-Naive and -Pretreated Patients

Characteristic	Ponatinib naive (n=21)	Ponatinib pretreated (n=31)
Time from diagnosis, median (range), years	1.8 (0.5-13.4)	5.2 (1.5-27.9)
No. of prior TKIs, n (%)		
1	9 (42.9)	0
2	9 (42.9)	7 (22.6)
3	3 (14.2)	15 (48.4)
≥4	0	9 (29.0)
BCR-ABL1^{IS} at screening, n (%)		
>0.1% to ≤1%	3 (14.2)	5 (16.1)
>1% to ≤10%	7 (33.3)	6 (19.4)
>10%	11 (52.5)	17 (54.8)
Atypical/unknown transcripts	0	3 (9.7)

Table 3. Patient Disposition

Patients, n (%)	N=52
Treatment ongoing	35 (67.3)
Treatment ended	17 (32.7)
Physician decision[a]	10 (19.2)
AE	4 (7.7)
Progressive disease[b]	3 (5.8)

[a] Most discontinuations due to physician decision were due to lack of efficacy (without satisfying the criteria for disease progression).
[b] Defined as progression to AP (n=2) or BP (n=1) according to European LeukemiaNet criteria.

- Patient disposition is presented in **Table 3**
 - Treatment was ongoing in 67.3% of patients at the time of data cutoff
 - Median duration of exposure was 68.4 weeks (range, 2-175 weeks)
 - Of 35 patients still on treatment, 31 (88.6%) received treatment for >48 weeks
 - Median dose intensity was 399.0 mg/day (range, 188-400 mg/day)
 - No patients died on treatment
- Overall MMR rate was 46.9% (**Table 4**)
 - Most patients who achieved MMR did so by 24 weeks
 - Median time to MMR was 12.1 weeks (range, 4 to 84 weeks)
- In the 23 of 49 patients who achieved MMR, *BCR-ABL1* levels at study entry were:
 - >0.1% to ≤1% in 6 patients
 - >1% to ≤10% in 10 patients
 - >10% in 7 patients
- 21 of 23 patients who achieved MMR maintained it to the cutoff
 - 1 patient who lost MMR subsequently regained it
- Kaplan-Meier–estimated rate of MMR duration at 144 weeks (2.8 years) was 87% (95% CI, 68.4%-100%)
- More ponatinib-naive patients achieved MMR than ponatinib-pretreated patients (**Figure 3**)

Table 4. MMR by Week

Patients, n (%)	n=49[a]
Overall MMR	23 (46.9)
MMR by time points	
24 Weeks	20 (40.8)
48 Weeks	21 (42.9)
72 Weeks	22 (44.9)
96 Weeks	23 (46.9)

[a] Of the 49 evaluable patients, 8 (15.4%), 13 (25.0%), and 28 (53.8%) had *BCR-ABL1*^{IS} levels >0.1% to ≤1%, >1% to ≤10%, and >10%, respectively, at screening.

Figure 3. MMR in Ponatinib-Naive and -Pretreated Patients[a]

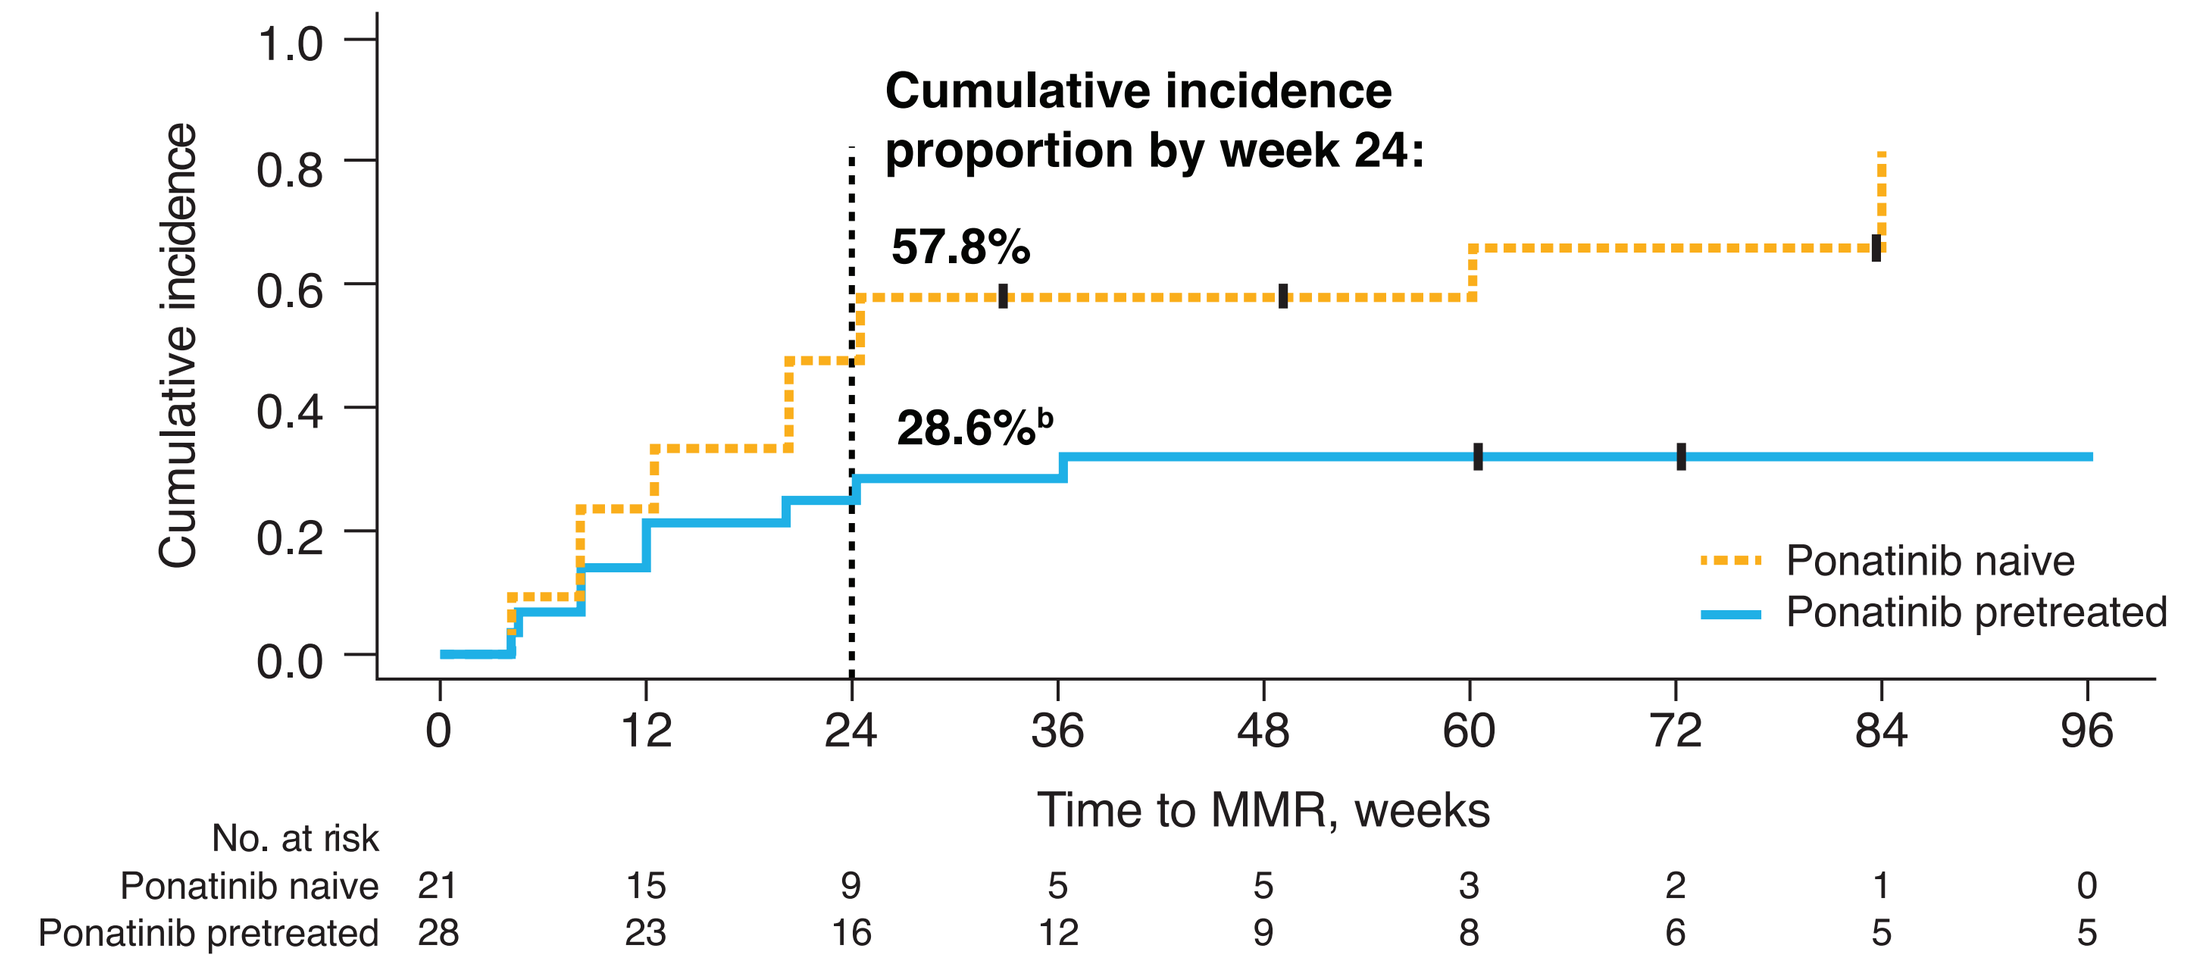
Cumulative incidence proportion by week 24:
57.8%
28.6%[b]
Cumulative incidence
1.0
0.8
0.6
0.4
0.2
0.0
0
12
24
36
48
60
72
84
96
Time to MMR, weeks
Ponatinib naive
Ponatinib pretreated
No. at risk
Ponatinib naive 21 15 9 5 5 3 2 1 0
Ponatinib pretreated 28 23 16 12 9 8 6 5 5

[a] Discontinuations and deaths were treated as competing risks. [b] Includes 5 patients who showed signs of resistance to ponatinib prior to study entry.

- Deeper molecular responses were achieved by both ponatinib-naive and -pretreated patients, although at higher rates in ponatinib-naive patients (**Table 5**)
 - In patients who achieved each level of response:
 - Median time to MR[4] was 20 weeks (range, 8-33 weeks)
 - Median time to MR[4.5] was 20 weeks (range, 8-49 weeks)
- Of the patients who did not achieve MMR, 3 had additional mutations at baseline besides T315I, and 6 acquired new mutations during the study (**Table 6**)
 - Of the 6 patients who acquired new mutations, 5 had been previously treated with ponatinib
- Among patients who lost MMR during the study, 1 had acquired a new mutation

Table 5. Cumulative MR[4] and MR[4.5] Rates in Evaluable Patients

Patients, n (%)	All patients (n=49)	Ponatinib naive (n=21)	Ponatinib pretreated (n=28)
MR[4]	13 (26.5)	8 (38.1)	5 (17.9)
MR[4.5]	10 (20.4)	7 (33.3)	3 (10.7)

MR[4], *BCR-ABL1*^{IS} ≤0.01%; MR[4.5], *BCR-ABL1*^{IS} ≤0.0032%.

Table 6. Additional *BCR-ABL1* Mutations[a]

Patients who did not achieve MMR	n=26
T315I with additional mutations at baseline [VAF], n	3
T315I/E255K [90/20]	1
T315I/F359I [80/30]	1
T315I/E355G [100/100]	1
Isolated T315I at baseline, n	23
No new mutations detected during the study, n	15
Mutations acquired during the study [VAF], n[b]	6
F359I [100], T315I/F359I [90/20]	1,1
A337T[c]/F359V [70/10]	1[d]
T315I/M351T [100/40]	1
T315I/M244V [60/10]	1
T315I/E453Q [100/30]	1[e]
Patients who lost MMR	**n=2**
T315I with additional mutations at baseline, n	0
Isolated T315I at baseline, n	2
No new mutations detected during the study, n	1[f]
Mutations acquired during the study [VAF], n[b]	1
F359V [80]	1

Ponatinib-naive patients; ponatinib-pretreated patients.
[a] Mutational status based on Sanger sequencing at central laboratory (measuring mutations between amino acids 35 and 510). Per protocol, mutational analysis was performed when there was unconfirmed loss of response or as needed; when a mutation was detected, additional mutational analyses were to be performed every 3 cycles.
[b] For mutations acquired during the study, the listed VAF is for the time of first detection of the mutation.
[c] Myristoyl pocket mutation.
[d] Patient progressed from CML-CP to CML-AP on study. Patient lost T315I and acquired A337T/F359V at the end of treatment assessment.
[e] Patient progressed from CML-AP to CML-BP on study. Patient maintained T315I and acquired E453Q in cycle 4 and discontinued 3 months later.
[f] The patient who lost MMR and did not acquire a mutation during the study eventually regained MMR.

- The most frequent AEs were fatigue, nausea, and diarrhea (**Table 7**)
- Other grade ≥3 AEs present in >1 patient: neutropenia (n=7); vomiting, alanine aminotransferase level increase, and hypertension (n=3 each); and anemia, γ-glutamyltransferase level increase, amylase level increase, peripheral edema, and pneumonia (n=2 each)
- 3 patients experienced ≥1 arterial occlusive event during this study (**Table 8**)
 - 3 of these events were serious, consisting of a grade 4 ischemic stroke, a grade 1 cerebrovascular accident, and a grade 3 peripheral vascular disorder

Table 7. Most Frequent On-Treatment AEs Regardless of Study Drug Relationship (Any Grade in ≥15% of Patients)

Preferred term, n (%)	All grades (N=52)	Grade ≥3 (N=52)
Any AE	52 (100)	30 (57.7)
Fatigue	14 (26.9)	1 (1.9)
Nausea	14 (26.9)	0
Diarrhea	11 (21.2)	1 (1.9)
Lipase level increased	11 (21.2)	8 (15.4)
Thrombocytopenia	10 (19.2)	9 (17.3)
Vomiting	10 (19.2)	3 (5.8)
Abdominal pain	8 (15.4)	4 (7.7)
Headache	8 (15.4)	1 (1.9)

Table 8. On-Treatment Arterial Occlusive AEs Regardless of Causality

Patients, n (%)	Patients, n (%) (N=52)	Grade	Serious	Time to onset, days
Any arterial occlusive AE	3 (5.8)			
Ischemic stroke[a]	1 (1.9)	4	Y	23
Cerebrovascular accident[b]	1 (1.9)	1	Y	140
Peripheral vascular disorder[c]	1 (1.9)	3	Y	505/602
Cerebral ischemia[a]	1 (1.9)	3	N	31

[a] Both events (ischemic stroke and cerebral ischemia) occurred in a single patient with ongoing relevant medical conditions (hypertension, ischemia, and arterial disorder) and worsening of the underlying disease.
[b] Occurred on the day after discontinuation for disease progression to CML-AP (ongoing relevant conditions: diabetes and hypertension).
[c] 2 episodes in the same patient, which started as serious events (requiring hospitalization) and continued as non serious. Possible contributing factors reported as prior TKI (past and ongoing relevant conditions: cerebrovascular accident and hypertension, respectively).

Acknowledgments

We thank the patients and their families and investigators and staff at participating study sites.
Editorial assistance was provided by Michelle Chadwick, PhD, (Nucleus Global) and was supported by Novartis Pharmaceuticals Corporation.

Disclosures

Cortes: Novartis, Takeda, Pfizer: consultancy, research funding; Sun Pharma: research funding. **Hughes:** Novartis, Bristol Myers Squibb: research funding, honoraria, board of directors or advisory committee; Novartis: speakers bureau. **Mauro:** Novartis, Bristol Myers Squibb, Pfizer, Takeda: consultancy, honoraria; Novartis, Bristol Myers Squibb, Sun Pharma: research funding. **Hochhaus:** Novartis, Takeda, Incyte, Bristol Myers Squibb, Pfizer: research funding. **Réa:** Novartis, Pfizer, Incyte: advisory boards; Novartis: steering committee; Pfizer, Incyte: educational activities. **Goh:** Pfizer, Johnson & Johnson, Amgen, MSD, Novartis, EUSA Pharma, Roche, Bristol Myers Squibb, AbbVie: honoraria. **Janssen:** Novartis, Bristol Myers Squibb: research funding; Incyte: speaker's fee; Novartis: other. **Heinrich:** Novartis, Deciphera, Blueprint Medicines: consultancy; Novartis: patents and royalties; Deciphera: speakers bureau. **Talpaz:** none. **Etienne:** Novartis, Bristol Myers Squibb, Pfizer, Incyte: consultancy, speakers bureau; Novartis, Bristol Myers Squibb: research funding. **Breccia:** Novartis, Incyte, Pfizer, Bristol Myers Squibb: consultancy, honoraria; AbbVie: consultancy. **Deininger:** Blueprint, Fusion Pharma, Medscape, Novartis, Sangamo, DisperSol: consultancy; Blueprint, Takeda, Novartis, SPARC, Leukemia & Lymphoma Society, Pfizer: research funding; Blueprint, Takeda, Sangamo: board of directors or advisory committee; Blueprint: other. **le Coutre:** Novartis, Pfizer, Incyte: honoraria. **Lang:** Novartis, Bristol Myers Squibb, Celgene, Incyte: consultancy, honoraria; Novartis: research funding. **Polydoros:** Novartis: employment, equity ownership. **Cacciatore:** Novartis: employment. **Stenson:** Novartis: employment. **Kim:** Novartis, Bristol Myers Squibb, Pfizer, IL-YANG: grants.

References

1. Hochhaus A, et al. *Leukemia.* 2020;34:966-984.
2. Byrgazov K, et al. *Haematologica.* 2018;103:e10-e12.
3. Manley PW, et al. *Leuk Res.* 2020;98:106458.
4. Wylie AA, et al. *Nature.* 2017;543:733-737.
5. Schoepfer J, et al. *J Med Chem.* 2018;61:8120-8135.
6. Hughes TP, et al. *N Engl J Med.* 2019;381:2315-2326.
7. Rea D, et al. *Blood.* 2018;132 (suppl 1). Abstract 792.