CML-375

Moshe Y. Levy, Moshe.Levy@BSWHealth.org

AIM4CML Trial in Progress: A Phase IIIb Study of Asciminib Monotherapy in Patients With Chronic Myeloid Leukemia in Chronic Phase With and Without the T315I Mutation

Moshe Y. Levy,[1,*] Michael J. Mauro,[2] Ghayas C. Issa,[3] David Andorsky,[4] Sarah Tomassetti,[5] Rodrigo Maegawa,[6] Alok Shrestha,[6] Michael Deininger[7]

[1]Baylor Scott & White Health, Dallas, TX, USA; [2]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [3]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [4]Rocky Mountain Cancer Centers, Boulder, CO, USA; [5]Harbor-UCLA Medical Center, West Carson, CA, USA; [6]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [7]The University of Utah, Salt Lake City, UT, USA

Scan to obtain:
- Poster
- Supplementary Podcast

http://novartis.medicalcongressposters.com/Default.aspx?doc=7a566

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

INTRODUCTION

- ATP-competitive TKIs that inhibit BCR-ABL1 are the standard of care for CML, and most patients with CML achieving optimal responses to treatment have near-normal life expectancies on TKI therapies[1-3]
- Many patients develop resistance to or intolerance of TKIs and are at risk of experiencing poor outcomes due to inadequate efficacy and/or safety of current TKIs[2,4-7]
 - Patients with treatment failure on or intolerance of successive lines of TKI therapy, including those who develop resistance mutations, have few remaining treatment options[2,6-8]
 - Patients harboring the T315I mutation are resistant to all approved TKIs, except for ponatinib[2,6]
- Asciminib, an investigational drug, is the first BCR-ABL1 inhibitor that Specifically Targets the ABL Myristoyl Pocket (STAMP) (**Figure 1**)[9-12]
- In phase I and III clinical trials, asciminib has demonstrated promising efficacy and safety in patients with CML-CP who are resistant to or intolerant of ≥2 prior TKIs[12,13]
- Here we describe the phase IIIb **A**sciminib **I**n **M**onotherapy **4**CML (AIM4CML) trial (ClinicalTrials.gov, NCT04666259), which has been initiated to reinforce existing data on the efficacy and safety of asciminib in this difficult-to-treat patient population

ASCIMINIB OVERVIEW

- Unlike approved TKIs for CML that bind to the ATP binding site of the BCR-ABL1 oncoprotein to inhibit its kinase activity, asciminib allosterically inhibits BCR-ABL1 via a novel mechanism of action (**Figure 1**)[9-11]
- Asciminib maintains activity against mutations that confer resistance to approved TKIs[9,10] and is not associated with toxicities resulting from off-target activity[11]
- In the phase III ASCEMBL study comparing asciminib with bosutinib in patients with CML-CP after ≥2 prior approved TKIs, asciminib demonstrated superior efficacy vs bosutinib and an improved safety profile[13]
 - The major molecular response (MMR) rate at week 24 was 25.5% with asciminib vs 13.2% with bosutinib
 - Fewer grade ≥3 adverse events (AEs) occurred with asciminib than with bosutinib (50.6% vs 60.5%) and fewer AEs leading to treatment discontinuation occurred with asciminib than with bosutinib (5.8% vs 21.1%)
- In a large phase I trial in heavily pretreated patients with CML, asciminib demonstrated efficacy and safety in patients with CML-CP with or without T315I mutations[12]
 - By 6 months, 37% of patients without and 25% of those with the T315I mutation achieved or maintained MMR
 - Major AEs included fatigue, headache, and increased lipase (occurring in ≥25% of patients)—most of which were grade 1 or 2

Figure 1. Asciminib Specifically Targets the ABL Myristoyl Pocket (STAMP)

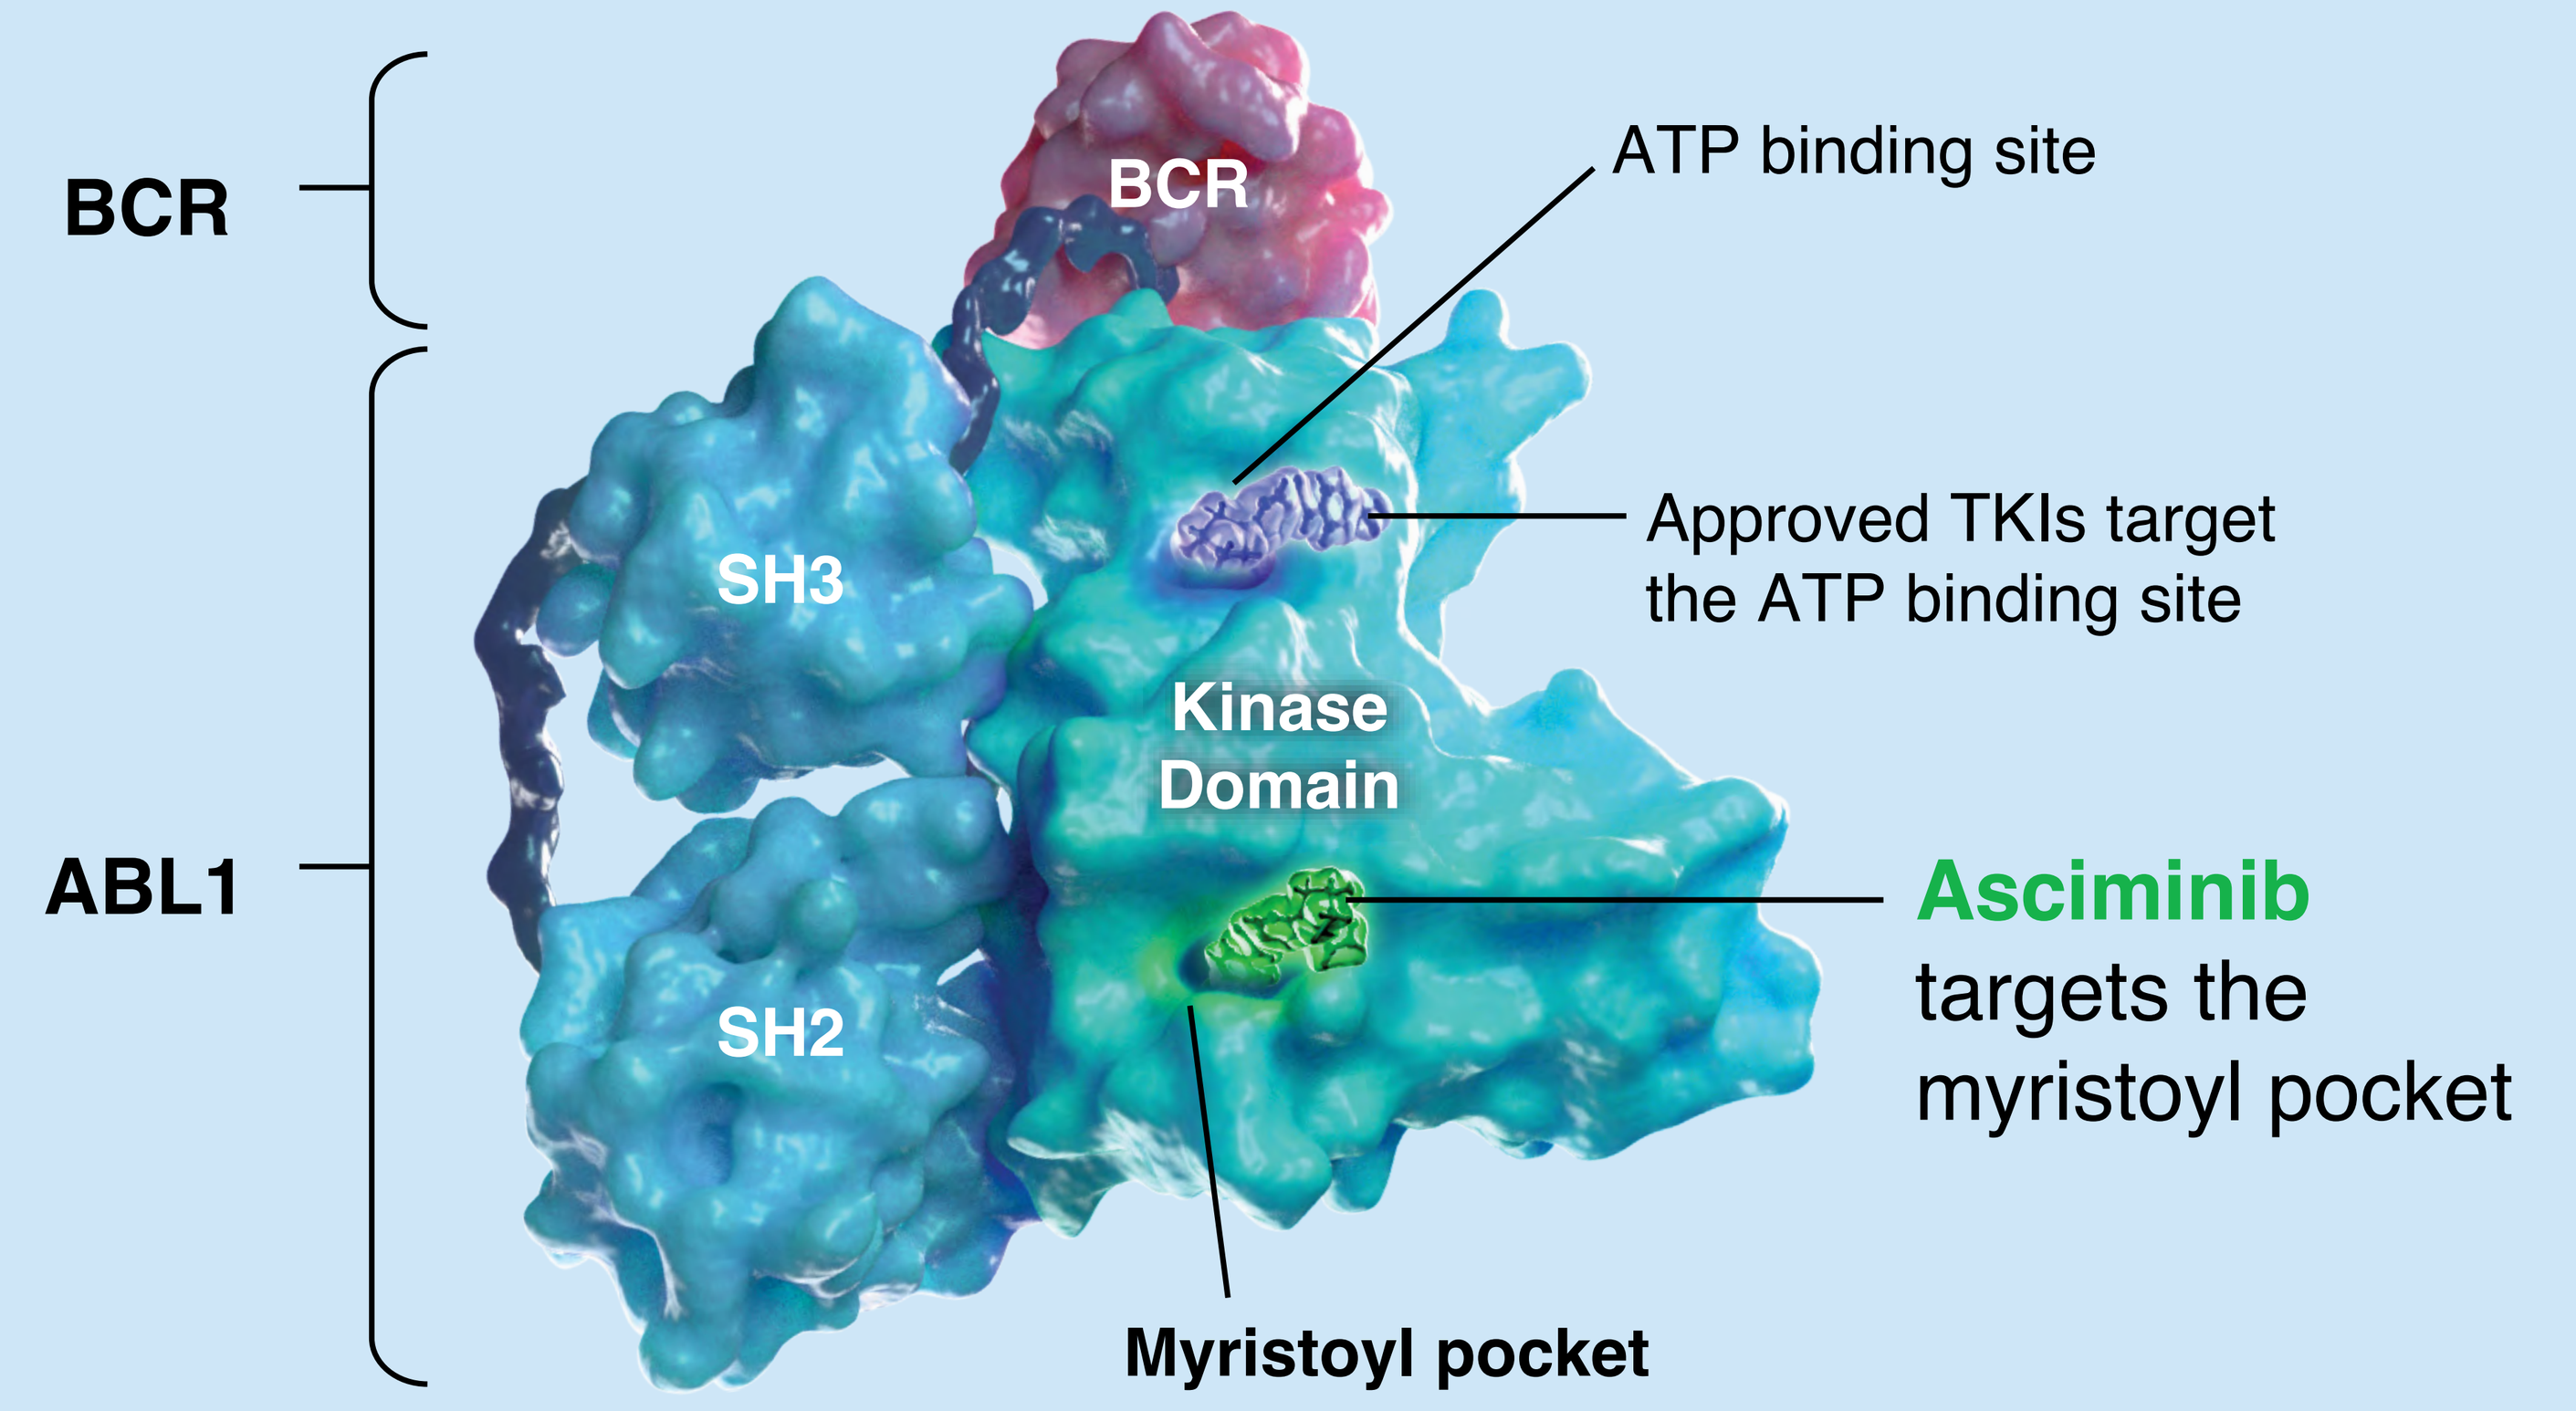
BCR
ABL1
BCR
SH3
SH2
Kinase Domain
ATP binding site
Approved TKIs target the ATP binding site
Asciminib targets the myristoyl pocket
Myristoyl pocket

Reprinted from Manley PW, et al. *Leuk Res.* 2020;98:106458, Copyright 2020, with permission from Elsevier.
ABL, Abelson tyrosine kinase; BCR, breakpoint cluster region; SH, Src homology domain.

STUDY DESIGN AND METHODS

- AIM4CML is a randomized, multicenter, phase IIIb, open-label, 3-cohort study of asciminib in patients with CML-CP without the T315I mutation after ≥2 prior TKIs and those with the T315I mutation after ≥1 prior TKI (**Figure 2**)
- Key eligibility criteria are summarized in **Table 1**
- Study objectives are described in **Table 2**

Figure 2. Study Design and Endpoints

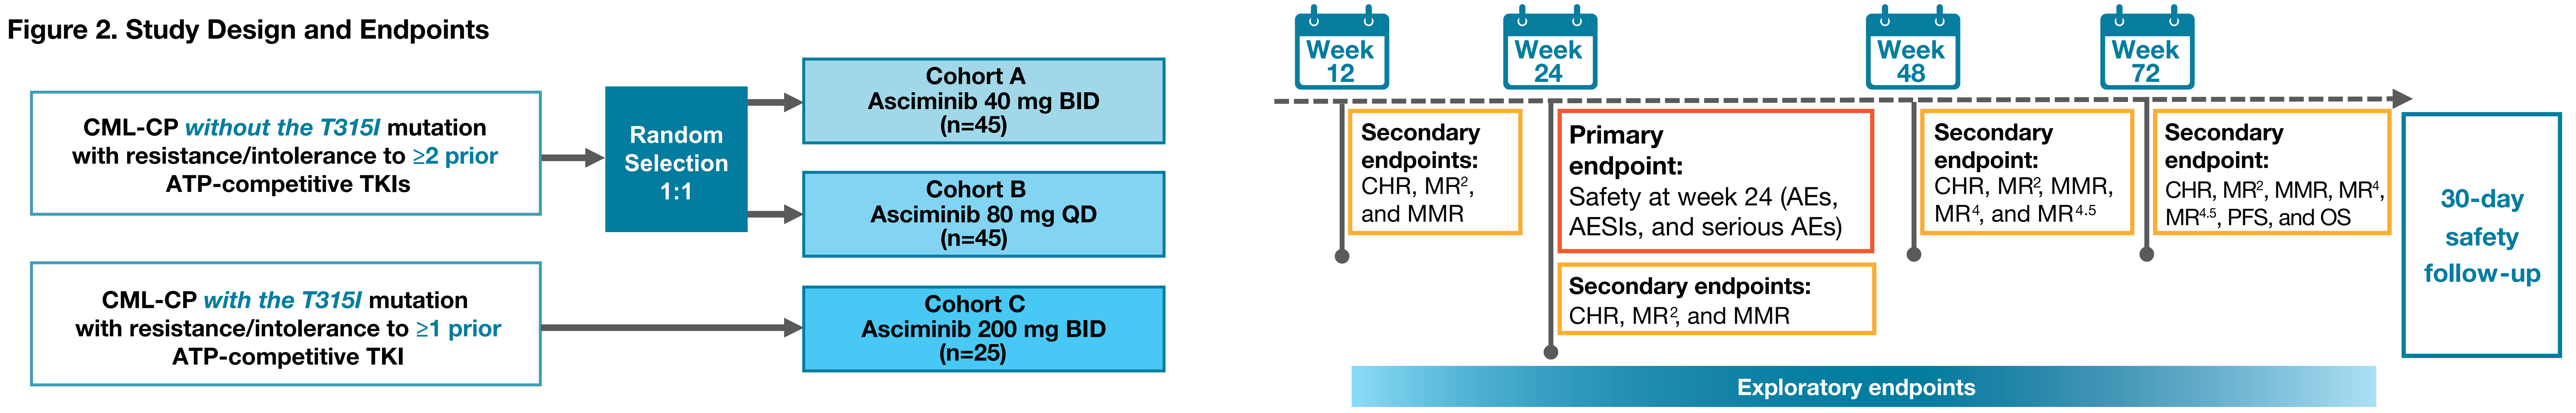
CML-CP without the T315I mutation with resistance/intolerance to ≥2 prior ATP-competitive TKIs
CML-CP with the T315I mutation with resistance/intolerance to ≥1 prior ATP-competitive TKI
Random Selection 1:1
Cohort A Asciminib 40 mg BID (n=45)
Cohort B Asciminib 80 mg QD (n=45)
Cohort C Asciminib 200 mg BID (n=25)
Week 12
Week 24
Week 48
Week 72
Secondary endpoints: CHR, MR², and MMR
Primary endpoint: Safety at week 24 (AEs, AESIs, and serious AEs)
Secondary endpoints: CHR, MR², and MMR
Secondary endpoint: CHR, MR², MMR, MR⁴, and MR4.5
Secondary endpoint: CHR, MR², MMR, MR⁴, MR4.5, PFS, and OS
30-day safety follow-up
Exploratory endpoints

AESIs, AEs of special interest; CHR, complete hematologic response; MR, molecular response; MR², *BCR-ABL1*[IS] ≤1%; MMR, *BCR-ABL1*[IS] ≤0.1%; MR⁴, *BCR-ABL1*[IS] ≤0.01%; MR4.5, *BCR-ABL1*[IS] ≤0.0032%, OS, overall survival; PFS, progression-free survival.

Table 1. Overview of Eligibility Criteria

Key Inclusion Criteria	Key Exclusion Criteria
Adults (≥18 years of age) with a diagnosis of CML-CP	Known second CML-CP after previous progression to CML-AP/BP
Prior treatment with ≥2 ATP-competitive TKIs in patients without the T315I mutation or ≥1 ATP-competitive TKI in patients with the T315I mutation	Previous hematopoietic stem cell transplant
Treatment failure (adapted from the 2020 ELN recommendations) or intolerance to the most recent TKI at screening	Cardiac or cardiac repolarization abnormality
Evidence of typical *BCR-ABL1* transcripts at screening	Severe and/or uncontrolled concurrent disease that could cause unacceptable safety risks or compromise compliance with the protocol
ECOG performance status of 0, 1, or 2	History of acute pancreatitis within 1 year of study entry or medical history of chronic pancreatitis
Acceptable laboratory values at screening, adequate end-organ function within 12 days of the first dose of asciminib (patients with mild to moderate renal and hepatic impairment may be eligible), and electrolytes within normal limits before the first dose of asciminib	Impaired GI function or GI disease that may significantly alter the absorption of study drug

AP, accelerated phase; BP, blast phase; ECOG, Eastern Cooperative Oncology Group; ELN, European LeukemiaNet; GI, gastrointestinal.

Table 2. Study Objectives

Primary Objective	• Evaluate the safety profile of asciminib monotherapy in patients in the 3L and beyond (3L+) in cohorts A and B
Secondary Objectives	• Evaluate the safety profile of asciminib monotherapy • Estimate hematologic response and MR rates by specific time points • Estimate deep MR rates by specific time points • Estimate time to and duration of hematologic response and MR • Assess rates of progression-free survival and overall survival during week 72
Exploratory Objectives	• Assess the pharmacokinetic profile of asciminib 40 mg BID, 80 mg QD, and 200 mg BID • Test the presence of low-level mutations during asciminib treatment • Determine the impact of asciminib on patient symptom burden and quality of life during week 72 • Evaluate health care resource utilization during week 72

3L, third line.

CONCLUSIONS

- Recruitment to the phase IIIb AIM4CML trial is underway. AIM4CML is evaluating the efficacy and safety of asciminib 40 mg twice daily (BID) and 80 mg once daily (QD) in patients with chronic myeloid leukemia in chronic phase (CML-CP) without the T315I mutation treated with ≥2 prior tyrosine kinase inhibitors (TKIs) and of asciminib 200 mg BID in patients with the T315I mutation treated with ≥1 prior TKI
- Approximately 115 patients with CML-CP (90 patients without and 25 patients with a T315I mutation) will be recruited across multiple sites in the United States
- The goal of this study is to reinforce the data with asciminib as ≥3rd line therapy reported in the phase III ASCEMBL study and to bolster efficacy and safety data in patients with T315I mutated CML-CP
- Asciminib, through its novel mechanism of action as the first BCR-ABL1 inhibitor Specifically Targeting the ABL Myristoyl Pocket (STAMP), has the potential to transform standard-of-care for patients in later lines of therapy with CML-CP who received ≥2 prior TKIs

This study is sponsored by Novartis Pharma AG.
Poster presented at: SOHO Annual Meeting; September 8-11, 2021; Houston, TX.

Acknowledgments

The authors thank all study participants and their families, the study investigators, and staff participating at the study sites.
Financial support for medical editorial assistance was provided by Novartis Pharmaceuticals Corporation. The authors thank Rohini Roy, PhD, of Scientific Pathways for medical editorial assistance with this poster presentation.

Disclosures

Levy (AbbVie: consultancy, speakers bureau; Amgen: consultancy, speakers bureau; AstraZeneca: consultancy, speakers bureau; Bristol Myers Squibb: consultancy, speakers bureau; Beigene: consultancy, speakers bureau; Gilead: consultancy, speakers bureau; Janssen: consultancy, speakers bureau; Jazz: consultancy, speakers bureau; Karyopharm: consultancy, speakers bureau; Morphosys: consultancy, speakers bureau; Seattle Genetics: consultancy, speakers bureau; Takeda: consultancy, speakers bureau; TG Therapeutics: consultancy, speakers bureau). **Mauro** (Bristol Myers Squibb: consultancy, research funding; Novartis: consultancy, research funding; Pfizer: research funding; Sun Pharma/SPARC: research funding; Takeda: research funding). **Issa** (Celgene: research funding; Kura Oncology: consultancy, research funding; Novartis: consultancy, research funding; Syndax: research funding). **Andorsky** (AbbVie: consultancy, research funding. AstraZeneca: research funding; Celgene: consultancy, research funding; Novartis: research funding). **Tomassetti** (Novartis: research funding). **Maegawa** (Novartis: employment). **Shrestha** (Novartis: employment). **Deininger** (Blueprint: consultancy, research funding, advisory boards; Fusion Pharma: consultancy; Takeda: consultancy, research funding, advisory boards; Novartis: consultancy, research funding; Incyte: consultancy, research funding, advisory boards; Sangamo: consultancy, advisory boards; SPARC: consultancy, research funding; Pfizer: consultancy, research funding; Medscape: consultancy; Dispersol: research funding; LLS: research funding.

References

1. Chopade P, Akard LP. *Clin Lymphoma Myeloma Leuk.* 2018;18:710-723. **2.** Hochhaus A, et al. *Leukemia.* 2020;34:966-984. **3.** Sasaki K, et al. *Lancet Haematol.* 2015;2:e186-e193. **4.** Jabbour E, et al. *Hematol Oncol Clin North Am.* 2011;25:981-995. **5.** Patel AB, et al. *Hematol Oncol Clin North Am.* 2017;31:589-612.**6.** NCCN Clinical Practice Guidelines in Oncology. Chronic Myeloid Leukemia V3.2021. **7.** Jabbour E, et al. *Clin Lymphoma Myeloma Leuk.* 2015;15:323-334. **8.** Akard LP. *Clin Adv Hematol Oncol.* 2013;11:421-432. **9.** Schoepfer J, et al. *J Med Chem.* 2018;61:8120-8135. **10.** Wylie AA, et al. *Nature.* 2017;543:733-737. **11.** Manley P, et al. *Leuk Res.* Published online September 29, 2020.doi:10.1016/j.leukres.2020.106458. **12.** Hughes TP, et al. *N Engl J Med.* 2019;381:2315-2326. **13.** Réa D, et al. *Blood.* Forthcoming 2021.