Poster CLL-354

Pooled Analysis of Cardiovascular Events from Clinical Trials Evaluating Acalabrutinib Monotherapy in Patients with Chronic Lymphocytic Leukemia (CLL)

Alessandra Ferrajoli,[1] John C. Byrd,[2] Paolo Ghia,[3] Jeff P. Sharman,[4] Peter Hillmen,[5] Deborah M. Stephens,[6] Clare Sun,[7] Wojciech Jurczak,[8] John M. Pagel,[9] Priti Patel,[10] Lin Tao,[10] Nataliya Kuptsova-Clarkson,[11] Javid Moslehi,[12] Richard R. Furman,[13] Jennifer R. Brown[14]

[1]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]The Ohio State University Comprehensive Cancer Center, Columbus, OH, USA; [3]Università Vita-Salute San Raffaele and IRCCS Ospedale San Raffaele, Milano, Italy; [4]Willamette Valley Cancer Institute/US Oncology, Eugene, OR, USA; [5]St. James's University Hospital, Leeds, United Kingdom; [6]University of Utah Huntsman Cancer Institute, Salt Lake City, UT, USA; [7]National Heart, Lung, and Blood Institute, Bethesda, MD, USA; [8]Maria Sklodowska-Curie National Research Institute of Oncology, Krakow, Poland; [9]Swedish Cancer Institute, Seattle, WA, USA; [10]AstraZeneca, South San Francisco, CA, USA; [11]AstraZeneca, Gaithersburg, MD, USA; [12]Vanderbilt University, Nashville, TN, USA; [13]Weill Cornell Medicine, New York Presbyterian Hospital, New York, NY, USA; [14]Dana-Farber Cancer Institute, Boston, MA, USA

Objective

- A retrospective pooled analysis of data from all clinical studies of acalabrutinib (from phase 1 to 3) in CLL was conducted to further characterize cardiac and hypertension-related effects of acalabrutinib in patients with CLL

Conclusions

- In this analysis, cardiac AEs occurred in 17% of patients with CLL treated with acalabrutinib monotherapy and rarely resulted in acalabrutinib discontinuation; 91% of patients with cardiac AEs had pre-existing risk factors
 - The rate of cardiac AE onset was generally constant over time on study
 - 25% of grade ≥3 cardiac AEs were reported during the first 6 months on acalabrutinib
- Atrial fibrillation incidence with acalabrutinib was 4% with a median follow-up duration of 25.9 months; for comparison, the incidence of new onset atrial fibrillation was 6% after a median follow-up of 7.3 years in a CLL population with no prior history of atrial fibrillation[1]
- Overall, these data suggest a low risk of cardiac AEs with acalabrutinib in patients with CLL
- These findings are supported by the results of the phase 3, randomized ELEVATE-RR trial (ACE-CL-006; NCT02477696), which showed a statistically lower incidence of any-grade atrial fibrillation and hypertension with acalabrutinib versus ibrutinib in patients with previously treated CLL with del(17p) or del(11q)[2]

Introduction

- Cardiovascular (CV) toxicities have been associated with the first-generation Bruton tyrosine kinase inhibitor (BTKi) ibrutinib, potentially due to off-target kinase inhibition[3]
 - For example, findings from an analysis of 562 patients with lymphoid malignancies suggested an increased incidence of hypertension and atrial fibrillation with ibrutinib treatment, with rates of 78% and 13%, respectively, reported after a median follow-up of 30 months[4]
- The next-generation BTKi acalabrutinib has greater selectivity for BTK than ibrutinib *in vitro* (**Figure 1**),[5,6] which may result in an improved safety profile[3,6-8]
- This analysis characterized CV adverse events (AEs) in patients with chronic lymphocytic leukemia (CLL) who received acalabrutinib monotherapy in clinical trials

Figure 1. Kinome profiling using KINOMEscan at a single dose (1 μM)[9]

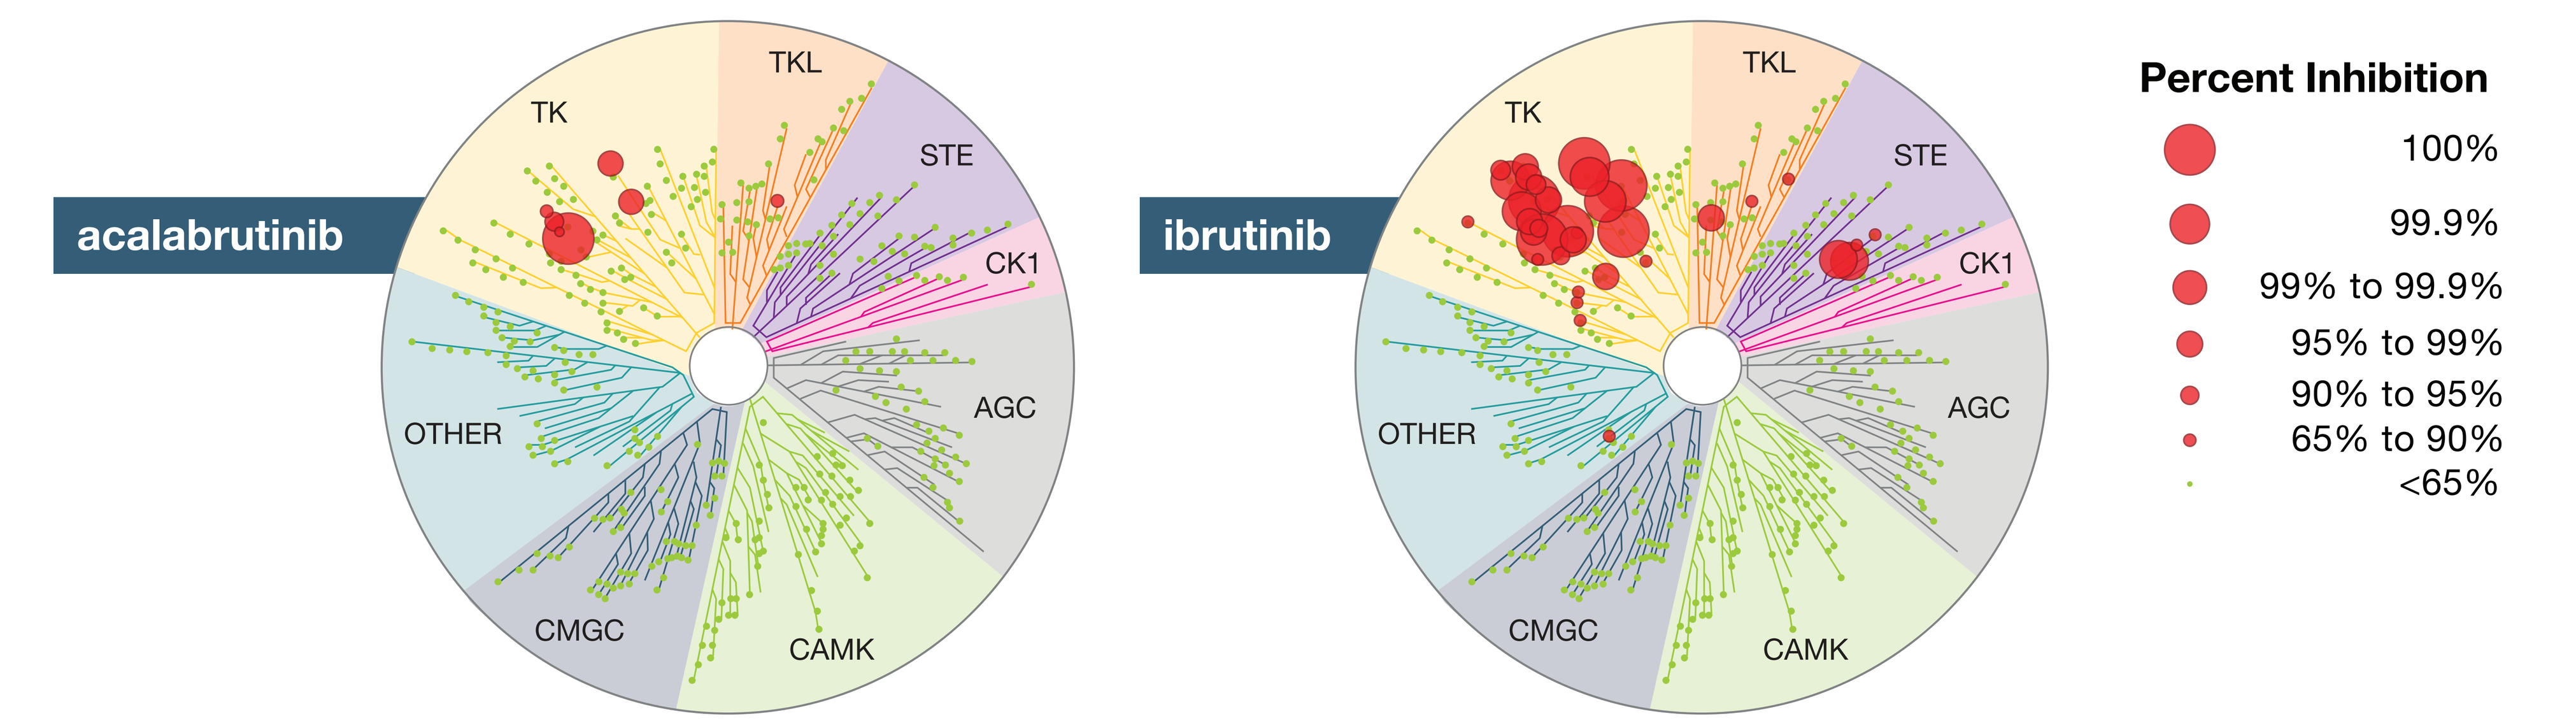
acalabrutinib
ibrutinib
TK
TKL
STE
CK1
AGC
CAMK
CMGC
OTHER
Percent Inhibition
100%
99.9%
99% to 99.9%
95% to 99%
90% to 95%
65% to 90%
<65%

Poster

Supplementary material

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials.

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from the authors of this poster.

Presented at the Annual Meeting of the Society of Hematologic Oncology (SOHO); September 8–11, 2021; Houston, Texas

Methods

- Data were included from a pooled population of patients treated with ≥1 dose of acalabrutinib monotherapy (N=762) (**Table 1**)
- Acalabrutinib was given orally at total daily doses of 100–400 mg and continued until progressive disease (PD) or toxicity
 - 78% of patients (n=596) received 100 mg twice daily (BID) dose only
 - 166 patients started on a different dose, of which 106 (14%) later switched to 100 mg BID
- Assessments:
 - Cardiac AEs (AEs categorized under the system organ class cardiac disorders)
 - Incidence, seriousness, severity, and relationship to acalabrutinib (per investigator assessment)
 - Time to onset and event duration
 - Event management and resolution (grade ≥3 cardiac AEs)
 - Incidence in first 6 months
 - Hypertension AEs (AEs categorized under the preferred term hypertension)
 - Incidence and time to onset
 - Cardiovascular and hypertension risk factors (based on patient medical histories)

Table 1. Pooled Studies

Study Name	Study Description	Number of Patients
ACE-CL-001 (NCT02029443)[6,10-12]	**Ph 1/2 study** of acalabrutinib in patients with CLL	301; TN/RR: 112/189
ACE-CL-007 (NCT02475681; **ELEVATE-TN**)[13a]	**Ph 3 study** of acalabrutinib ± O vs C+O in TN CLL	224; all TN
ACE-CL-309 (NCT02970318; **ASCEND**)[14]	**Ph 3 study** of acalabrutinib vs IdR or BR in RR CLL	189; all RR
15-H-0016 (NCT02337829)[15]	**Ph 2 study** of acalabrutinib in patients with RR or TN with del(17p) CLL	48; TN/RR: 16/32

BR, bendamustine plus rituximab; C, chlorambucil; IdR, idelalisib plus rituximab; O, obinutuzumab; Ph, phase; RR, relapsed/refractory; TN, treatment-naïve.
[a]Study has acalabrutinib monotherapy and combination therapy arms; only monotherapy patients were included.

Results

Patients, Exposure, and Disposition

- Patient demographics and baseline characteristics were similar between the total acalabrutinib population and the subgroup of patients with cardiac AEs (**Table 2**)
- Among all patients (N=762), median follow-up duration was 25.9 months (range, 0.0–58.5; n=761)
- Most patients (73%; n=553) remained on acalabrutinib at data cutoff
 - 208 patients (27%) discontinued acalabrutinib, most commonly due to PD (11%; n=82) or AEs (9%; n=70)
- 129 patients (17%) experienced cardiac AEs of any grade while on study; cardiac AEs led to acalabrutinib discontinuation in 7 patients (0.9%)
- 91% (n=117/129) of patients with cardiac AEs vs 80% (n=503/633) without cardiac AEs had ≥1 CV risk factor before starting acalabrutinib
 - The most prevalent CV risk factors (defined in Supplemental Methods) for patients with vs without cardiac AEs were hypertension (65% [n=84] vs 53% [n=333]), diabetes mellitus (19% [n=24] vs 12% [n=79]), hyperlipidemia (16% [n=21] vs 11% [n=67]), hypothyroidism (16% [n=21] vs 12% [n=73]), hypercholesterolemia (14% [n=18] vs 16% [n=104]), and atrial fibrillation (12% [n=16] vs 6% [n=40])

Table 2. Patient Demographics and Baseline Characteristics

Demographic/Baseline Characteristic	All Patients (N=762)	Patients With Cardiac Events (n=129)
Age, median (range), years	67.0 (32–89)	69.0 (42–84)
Male, n (%)	508 (67)	83 (64)
ECOG PS score ≤1, n (%)	707 (93)	122 (95)
Number of prior regimens, median (range)	1 (0–13)	1 (0–10)
Treatment-naïve disease, n (%)	352 (46)	58 (45)
Relapsed/refractory disease, n (%)	410 (54)	71 (55)

ECOG PS, Eastern Cooperative Oncology Group performance status.

Safety

- The most frequent any grade cardiac events were atrial fibrillation, palpitations, and tachycardia (**Table 3**)
- Among the 37 patients with grade ≥3 cardiac AEs, 18 (49%) were continuing acalabrutinib at data cutoff
 - 6 of the 37 patients (16%) discontinued acalabrutinib due to those grade ≥3 cardiac AEs (acute myocardial infarction [n=2], cardiac failure congestive [n=2], cardiac failure [n=1], cardiac tamponade [n=1])
 - 13 patients discontinued acalabrutinib due to other AEs (n=4), PD (n=5), death (n=3), and other reasons (n=1)

Results continued

Table 3. Incidence of Cardiac Adverse Events

	All Patients (N=762)	
Event	Any Grade	Grade ≥3[c]
Any cardiac AE,[a] n (%),[b] [number of events]	129 (17), [199]	37 (5), [51]
Most common cardiac AEs (preferred terms; occurring in ≥4 patients), n (%), [number of events]		
Atrial fibrillation/flutter[d]	38 (5), [48]	11 (1), [12]
Atrial fibrillation	34 (4), [44]	10 (1), [11]
Atrial flutter	4 (0.5), [4]	1 (0.1), [1]
Palpitations	23 (3), [27]	0
Tachycardia	17[e] (2), [18]	0
Sinus tachycardia	11[e] (1), [13]	1 (0.1), [1]
Angina pectoris	10 (1), [11]	2 (0.3), [2]
Bradycardia	9 (1), [10]	2 (0.3), [2]
Cardiac failure	6 (0.8), [6]	3 (0.4), [3]
Acute myocardial infarction	5 (0.7), [6]	5 (0.7), [6]
Supraventricular tachycardia	4[e] (0.5), [4]	1 (0.1), [1]

[a]AEs categorized under the system organ class cardiac disorders.
[b]199 AEs were reported in 129 patients (17%). No events under the preferred terms "sudden death" or "sudden cardiac death" were reported.
[c]Other grade ≥3 AEs of interest included complete AV block (n=2; 0.3%), acute coronary syndrome (n=1; 0.1%), second-degree AV block (n=1; 0.1%), and ventricular fibrillation (n=1; 0.1%).
[d]Patients with "atrial fibrillation" or "atrial flutter" preferred terms combined. There was no overlap between patients with "atrial fibrillation" and "atrial flutter" events.
[e]One patient had both "tachycardia" and "sinus tachycardia" events. Another patient had both "sinus tachycardia" and "supraventricular tachycardia" events. A third patient had both "tachycardia" and "supraventricular tachycardia" events. All other reports of "tachycardia," "sinus tachycardia," and "supraventricular tachycardia" occurred in unique patients.
AE, adverse event; AV, atrioventricular.

- 51 grade ≥3 cardiac events were reported in 37 patients (grade 3=37; grade 4=12; grade 5=2)
 - Grade 5 events included acute myocardial infarction (patient had preexisting hypertension, type 2 diabetes mellitus, cardiovascular disorder, and chronic kidney disease) and congestive heart failure (patient had prior history of congestive cardiac failure, aortic stenosis, and atrial fibrillation)
 - 13 of the 51 events (in 9 patients) occurred in the first 6 months on acalabrutinib (**Supplemental Table 1**)
- Among the 51 grade ≥3 events:
 - 16 (31%) led to dose delay and 6 (12%) led to acalabrutinib discontinuation
 - 36 (71%) were managed with concomitant meds, and 43 (84%) resolved
 - Of the events that resolved, 15 were associated with dose delays, 4 resulted in drug withdrawal, and 24 had no associated dose modifications or changes
- Time to onset and duration of events are reported for cardiac events overall in **Figure 2A** and for atrial fibrillation/flutter and hypertension in **Figure 2B**
- Among 38 patients with atrial fibrillation/flutter, 7 (18%) had a prior history of arrhythmia/atrial fibrillation/flutter
- Among 67 patients with hypertension events, 43 (64%) had pre-existing hypertension
 - Other than pre-existing hypertension, the most common risk factors (incidence >5%) included hyperlipidemia (19% [n=13]), hypercholesterolemia (19% [n=13]), diabetes mellitus (10% [n=7]), coronary artery disease (9% [n=6]), chronic obstructive pulmonary disease (7% [n=5]), and coronary artery bypass (6% [n=4])

Figure 2. Time to Onset of (A) Cardiac Events[a] and (B) Atrial Fibrillation/Flutter and Hypertension

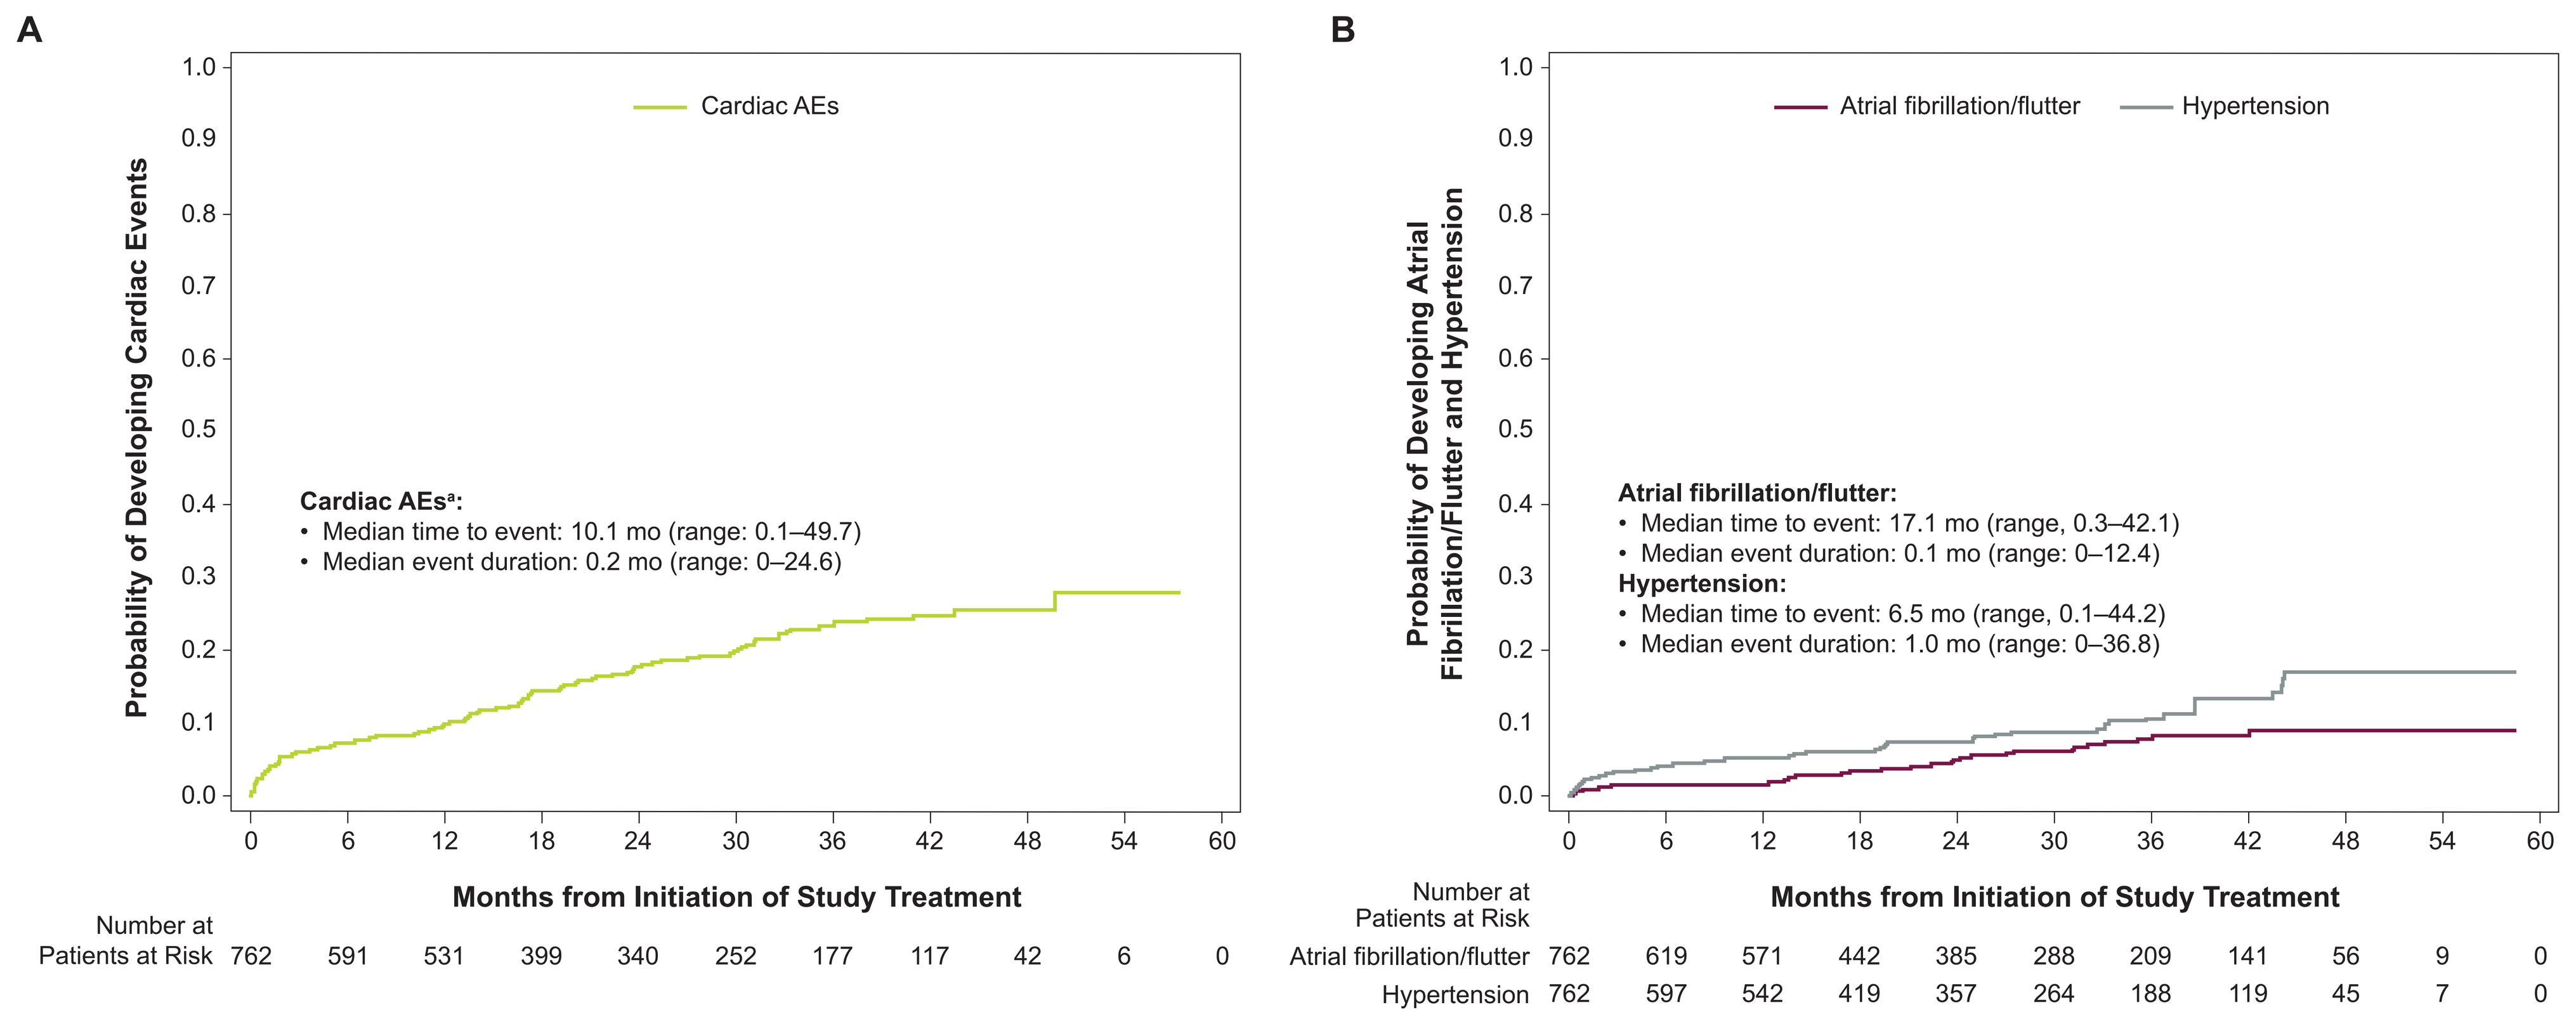
A
Cardiac AEs
Probability of Developing Cardiac Events
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
Cardiac AEs[a]:
• Median time to event: 10.1 mo (range: 0.1–49.7)
• Median event duration: 0.2 mo (range: 0–24.6)
0 6 12 18 24 30 36 42 48 54 60
Months from Initiation of Study Treatment
Number at Patients at Risk 762 591 531 399 340 252 177 117 42 6 0
B
Atrial fibrillation/flutter
Hypertension
Probability of Developing Atrial Fibrillation/Flutter and Hypertension
Atrial fibrillation/flutter:
• Median time to event: 17.1 mo (range, 0.3–42.1)
• Median event duration: 0.1 mo (range: 0–12.4)
Hypertension:
• Median time to event: 6.5 mo (range, 0.1–44.2)
• Median event duration: 1.0 mo (range: 0–36.8)
0 6 12 18 24 30 36 42 48 54 60
Months from Initiation of Study Treatment
Number at Patients at Risk
Atrial fibrillation/flutter 762 619 571 442 385 288 209 141 56 9 0
Hypertension 762 597 542 419 357 264 188 119 45 7 0

[a]AEs categorized under the system organ class cardiac disorders.

References

1. Shanafelt TD, et al. *Leuk Lymphoma*. 2017;58:1630. **2.** Byrd JC, et al. *J Clin Oncol*. 2021. https://doi.org/10.1200/JCO.21.01210. **3.** Pal Singh S, et al. *Mol Cancer*. 2018;17:57. **4.** Dickerson T, et al. *Blood*. 2019;134:1919-28. **5.** Barf T, et al. *J Pharmacol Exp Ther*. 2017;363:240-52. **6.** Byrd JC, et al. *N Engl J Med*. 2016;374:323-32. **7.** Byrd JC, et al. *N Engl J Med*. 2013;369:32-42. **8.** Byrd JC, et al. *N Engl J Med*. 2014;371:213-23. **9.** Kaptein A, et al. *Blood*. 2018;132(Suppl 1):1871. **10.** Byrd JC, et al. *Blood*. 2020;135:1204-13. **11.** Awan FT, et al. *Blood Adv*. 2019;3:1553-62. **12.** Byrd JC, et al. *Blood*. 2018;132(Suppl 1):692. **13.** Sharman JP, et al. *Lancet*. 2020;395:1278-91. **14.** Ghia P, et al. *J Clin Oncol*. 2020;38:2849-61. **15.** Sun C, et al. *Blood*. 2020;136:93-105.

Acknowledgments

We thank all the patients, their families, and the investigators and their study teams who participated in this study. This study was sponsored by Acerta Pharma, South San Francisco, CA, a member of the AstraZeneca Group. Medical writing support was provided by Peloton Advantage, LLC (Parsippany, NJ, USA), an OPEN Health company and funded by Acerta Pharma, a member of the AstraZeneca Group.

Disclosures

A Ferrajoli: Nothing to disclose. **JC Byrd:** Research Funding – Acerta Pharma, Syndax, Vincera, Novartis, Kartos Therapeutics, Trillium, Pharmacyclics LLC, Gilead, TG Therapeutics, BeiGene; Consultancy – Janssen. Speakers Bureau – Pharmacyclics LLC, Gilead, TG Therapeutics, Novartis, Janssen. Other – Leukemia and Lymphoma Society, Pharmacyclics LLC, Janssen, Novartis, Gilead, TG Therapeutics. **P Ghia:** Consultancy, Honoraria – AbbVie, Adaptive, ArQule/MSK, Acerta/AstraZeneca, BeiGene, Celgene/Juno, Janssen, Lilly/Loxo, MEI. Research Funding – AbbVie, Janssen, Gilead, Novartis, Sunesis. **JP Sharman:** Consultancy, Research Funding – Genentech, AstraZeneca, Pharmacyclics, Pfizer, AbbVie, TG Therapeutics, Acerta, Roche, Celgene, Bristol Myers Squibb. Research Funding – BeiGene. **P Hillmen:** Travel, Accommodations, Expenses – AbbVie, Janssen. Honoraria, Research Funding – F. Hoffmann-LaRoche. Honoraria – AstraZeneca. Research Funding – Pharmacyclics, Gilead. Employment – University of Leeds. **DM Stephens:** Consultancy – Janssen, Pharmacyclics, Innate, BeiGene. Consultancy, Research Funding – Karyopharm. Research Funding – Gilead, Verastem, Juno, Arqule, Acerta, MingSight. **C Sun:** Research Funding – Genmab. **W Jurczak:** Consultancy, Current Employment – Maria Sklodowska-Curie National Research Institute of Oncology. Research Funding, Previous Employment – Jagiellonian University. Research Funding – Janssen, MeiPharma, Merck, Pharmacyclics, Roche, Takeda, TG Therapeutics. **JM Pagel:** Consultancy – TG Therapeutics, AstraZeneca, BeiGene, Loxo Oncology. **P Patel:** Employment, Stockholder – AstraZeneca. **L Tao:** Employment, Stockholder – AstraZeneca. **N Kuptsova-Clarkson:** Employment – AstraZeneca. **J Moslehi:** Consultancy – AstraZeneca, Janssen, Bristol Myers Squibb, Boston Biomedical, Immunocure, Myovant, Deciphera. **RR Furman:** Consultancy – AbbVie, Acerta, Beigene, Genentech, Incyte, Loxo Oncology, Oncotarget, Pharmacyclics, Sunesis, Verastem. Consultancy, Research Funding – AstraZeneca, TG Therapeutics. Consultancy, Speakers Bureau – Janssen. **JR Brown:** Consultancy – Abbvie, AstraZeneca, Beigene, Catapult, Dynamo Therapeutics, Eli Lilly and Company, Juno/Celgene, Kite, MEI Pharma, Nextcea, Novartis, Octapharma, Pfizer, Rigel Pharm; Board of Directors, Advisory Committee – Invectys; Research Funding – Gilead, Loxo, Sun, Verastem.