Poster CLL-115

First Results of a Head-to-Head Trial of Acalabrutinib versus Ibrutinib in Previously Treated Chronic Lymphocytic Leukemia

John Byrd,[1] Peter Hillmen,[2] Paolo Ghia,[3] Arnon P. Kater,[4] Asher Chanan-Khan,[5] Richard R. Furman,[6] Susan O'Brien,[7] Mustafa Nuri Yenerel,[8] Arpad Illés,[9] Neil Kay,[10] Jose A. Garcia-Marco,[11] Anthony Mato,[12] Javier Pinilla-Ibarz,[13] John F. Seymour,[14] Stephane Lepretre,[15] Stephan Stilgenbauer,[16] Tadeusz Robak,[17] Priti Patel,[18] Kara Higgins,[18] Sophia Sohoni,[18] Wojciech Jurczak[19]

[1]The Ohio State University Comprehensive Cancer Center, Columbus, OH, USA; [2]St. James's University Hospital, Leeds, UK; [3]Università Vita-Salute San Raffaele and IRCCS Ospedale, San Raffaele, Milano, Italy; [4]Amsterdam University Medical Centers, Amsterdam, on behalf of Hovon, Netherlands; [5]Mayo Clinic Jacksonville, Jacksonville, FL, USA; [6]Weill Cornell Medicine, New York Presbyterian Hospital, New York, NY, USA; [7]Chao Family Comprehensive Cancer Center, University of California-Irvine, Irvine, CA, USA; [8]Istanbul University, Istanbul Faculty of Medicine, Istanbul, Turkey; [9]University of Debrecen, Debrecen, Hungary; [10]Mayo Clinic Rochester, Rochester, MN, USA; [11]Hospital Universitario Puerta de Hierro-Majadahonda, Madrid, Spain; [12]University of Pennsylvania, Philadelphia, PA, USA; [13]Moffitt Cancer Center, Tampa, FL, USA; [14]Peter MacCallum Cancer Centre & Royal Melbourne Hospital, Victoria, Australia; [15]Centre Henri Becquerel and Normandie University UNIROUEN, Rouen, France; [16]University of Ulm, Ulm, Germany; [17]Medican University of Lodz, Lodz, Poland; [18]AstraZeneca, South San Francisco, CA, USA; [19]Maria Sklodowska-Curie National Research Institute of Oncology, Krakow, Poland

Objective

- To report efficacy and safety results from the first head-to-head trial (ELEVATE-RR) comparing acalabrutinib and ibrutinib in patients with previously treated chronic lymphocytic leukemia (CLL) and del(17p) or del(11q)

Conclusions

- Acalabrutinib was non-inferior to ibrutinib on the primary endpoint of independent review committee (IRC)-assessed progression-free survival (PFS) (hazard ratio [HR]: 1.00 [95% CI: 0.79, 1.27])
- Acalabrutinib demonstrated lower frequencies of common adverse events (AEs), grade ≥3 AEs, serious AEs (SAEs), and treatment discontinuations due to AEs overall
- Cardiovascular events were less common with acalabrutinib vs ibrutinib
 - Afib/flutter events (any grade) were significantly less frequent with acalabrutinib vs ibrutinib (9.4% vs 16%; *P*=0.02)
 - Hypertension also was less frequent with acalabrutinib
- Other commonly reported AEs, including diarrhea, arthralgia, and bruising (any-grade bleeding) events, were also less frequent with acalabrutinib vs ibrutinib
- These results demonstrate that acalabrutinib is better tolerated and has similar efficacy to ibrutinib in patients with previously treated CLL

Introduction

- Bruton tyrosine kinase (BTK) is critical for CLL cell proliferation and survival[1-3]
- Ibrutinib was the first irreversible BTK inhibitor[4] approved for adults with CLL/small lymphocytic lymphoma (SLL)[5]
 - Ibrutinib treatment is associated with AEs, particularly cardiovascular toxicities, that can lead to treatment discontinuation[6-8]
 - Ibrutinib also binds to non-BTK kinases,[4,9,10] likely contributing to ibrutinib-associated AEs[4,10-12]
- Acalabrutinib is a next-generation, more selective, irreversible BTK inhibitor approved for CLL/SLL[13] with decreased alternative-target activity vs ibrutinib in vitro,[4,9] potentially resulting in an improved tolerability profile
- We report the results from the first head-to-head trial (ELEVATE-RR) comparing the safety and efficacy of acalabrutinib and ibrutinib in patients with previously treated CLL and del(17p) or del(11q)

Methods

- This phase 3, randomized, multicenter, open-label, non-inferiority trial included patients with CLL who had received ≥1 prior therapies and had del(17p) and/or del(11q) (**Figure 1**)
 - Patients received oral acalabrutinib 100 mg twice daily (BID) or oral ibrutinib 420 mg once daily (QD) until disease progression or unacceptable toxicity
 - Randomization was stratified by del(17p) status, Eastern Cooperative Oncology Group (ECOG) performance status, and number of prior therapies
- The primary study endpoint was non-inferiority on IRC-assessed PFS (**Figures 1 and 2**)
- If non-inferiority on the primary endpoint was met, superiority of acalabrutinib on the secondary endpoints was tested in the following prespecified order: any-grade atrial fibrillation/flutter (afib/flutter), grade ≥3 infection, Richter transformation, and overall survival (OS) (**Figure 2**)

Poster

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials.

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from the authors of this poster.

Presented at the Annual Meeting of the Society of Hematologic Oncology (SOHO); September 8–11, 2021; Houston, Texas

Methods (cont'd)

Figure 1. ELEVATE-RR Study Design

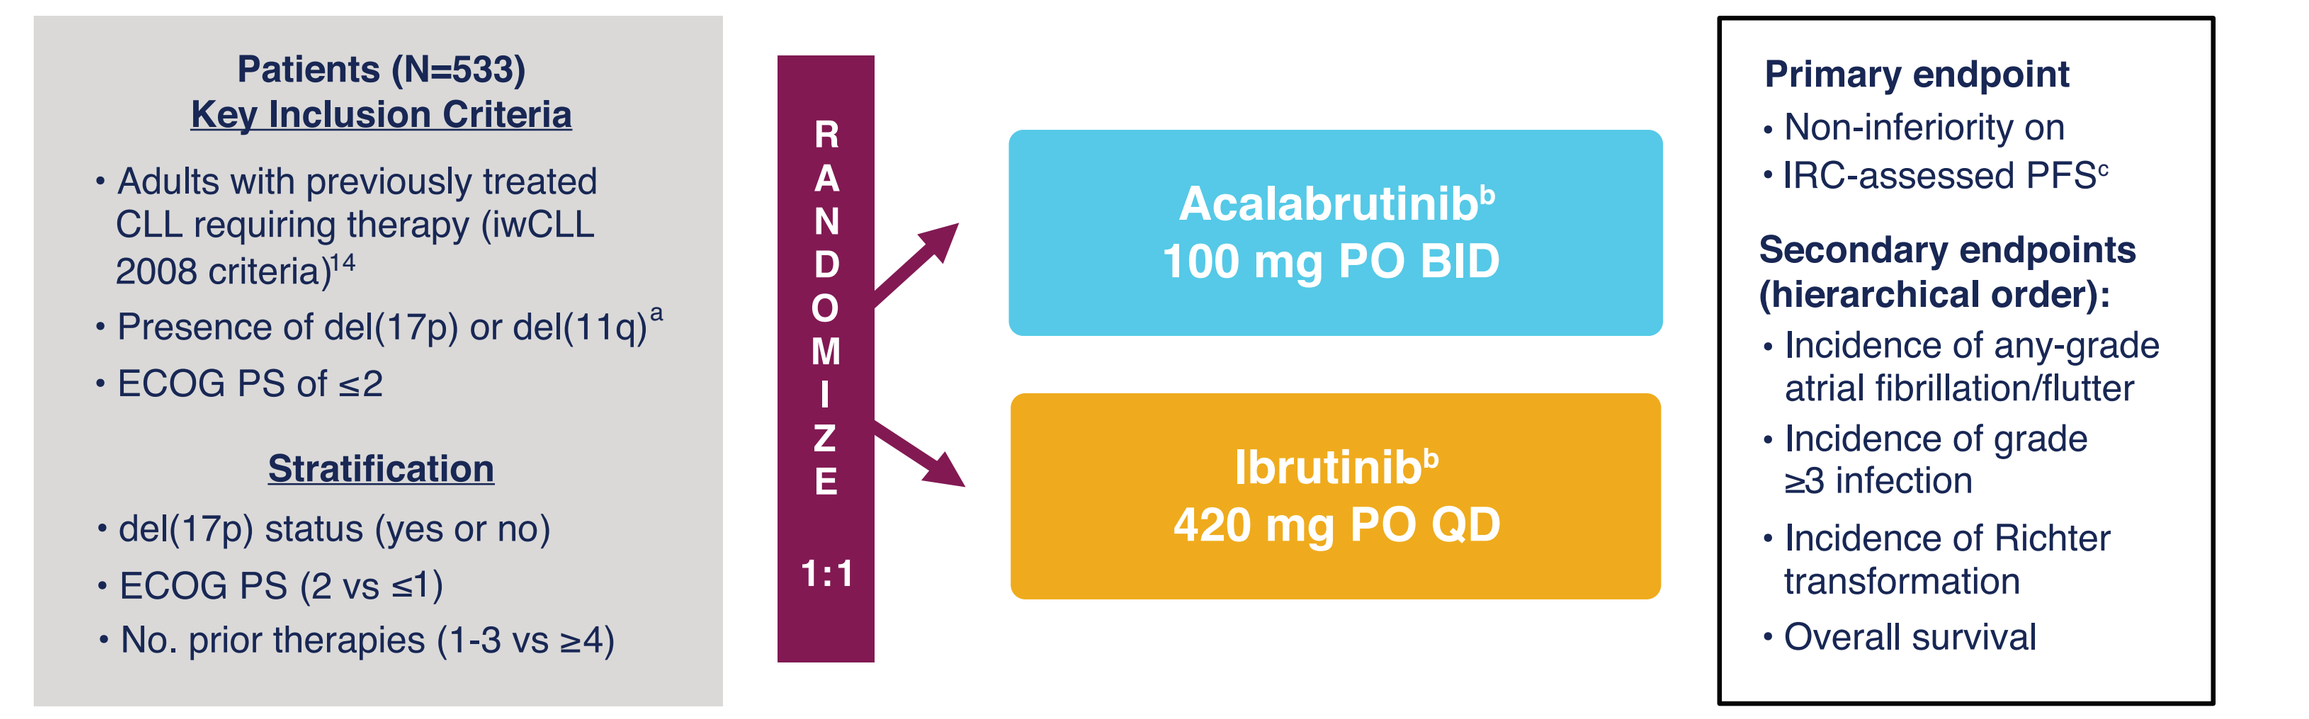
Patients (N=533)
Key Inclusion Criteria
• Adults with previously treated CLL requiring therapy (iwCLL 2008 criteria)14
• Presence of del(17p) or del(11q)a
• ECOG PS of ≤2
Stratification
• del(17p) status (yes or no)
• ECOG PS (2 vs ≤1)
• No. prior therapies (1-3 vs ≥4)
RANDOMIZE
1:1
Acalabrutinibb 100 mg PO BID
Ibrutinibb 420 mg PO QD
Primary endpoint
• Non-inferiority on
• IRC-assessed PFSc
Secondary endpoints (hierarchical order):
• Incidence of any-grade atrial fibrillation/flutter
• Incidence of grade ≥3 infection
• Incidence of Richter transformation
• Overall survival

- Key exclusion criteria: significant cardiovascular disease; concomitant treatment with warfarin or equivalent vitamin K antagonist; prior treatment with ibrutinib, a BCR inhibitor (eg, BTK , PI3K, or Syk inhibitors), or a BCL-2 inhibitor (eg, venetoclax)

NCT02477696 (ACE-CL-006).
[a]By central laboratory testing; [b]continued until disease progression or unacceptable toxicity; [c]conducted after enrollment completion and accrual of ~250 IRC-assessed PFS events. BCL-2, B-cell leukemia/lymphoma-2; BCR, B-cell receptor; BID, twice daily; BTK, Bruton tyrosine kinase; CLL, chronic lymphocytic leukemia; ECOG PS, Eastern Cooperative Oncology Group performance status; IRC, independent review committee; iwCLL, International Workshop on CLL; PFS, progression-free survival; PI3K, phosphatidylinositol 3-kinase; PO, orally; QD, once daily.

Figure 2. Hierarchical Analysis

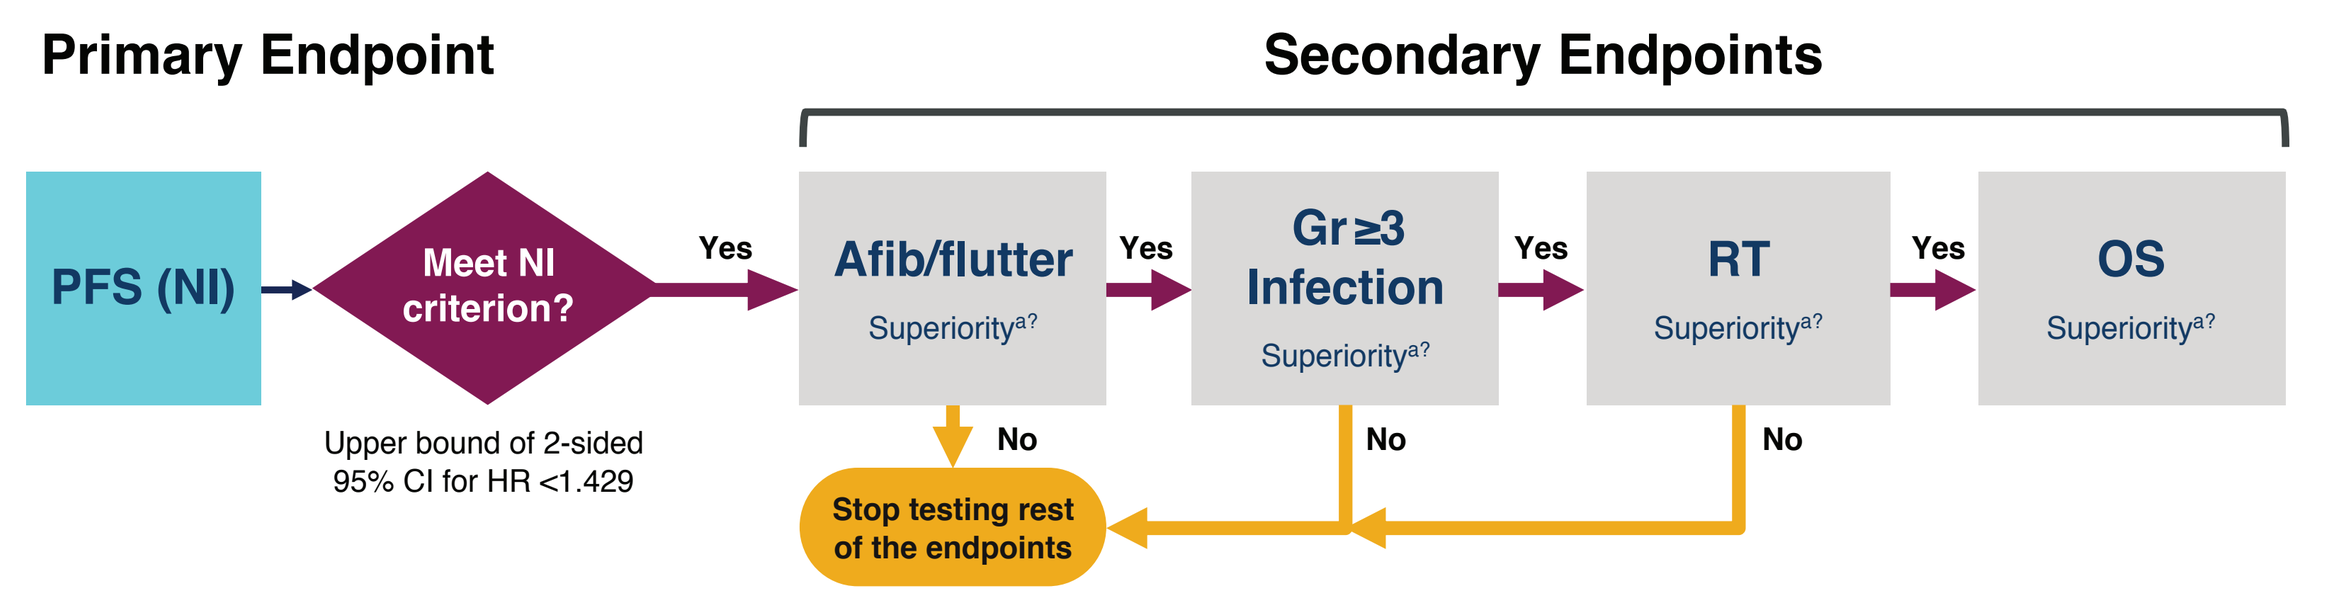
Primary Endpoint
Secondary Endpoints
PFS (NI)
Meet NI criterion?
Upper bound of 2-sided 95% CI for HR <1.429
Yes
Afib/flutter
Superioritya?
Yes
Gr≥3 Infection
Superioritya?
Yes
RT
Superioritya?
Yes
OS
Superioritya?
No
No
No
Stop testing rest of the endpoints

NI on the primary endpoint (IRC-assessed PFS) assessed using a Cox proportional-hazards model stratified by del(17p) status and no. prior therapies (1–3 vs ≥4). Superiority on secondary endpoints was tested hierarchically at a 2-sided 0.05 significance level: [a]Afib/flutter, Gr≥3 infection, and RT: CMH test adjusted for del(17p) status and no. prior therapies (1–3 vs ≥4); [b]OS: KM methods and stratified log-rank test.
Afib/flutter, atrial fibrillation/flutter; CI, confidence interval; CMH, Cochran-Mantel-Haenszel; Gr, grade; HR, hazard ratio; IRC, independent review committee; KM, Kaplan-Meier; NI, non-inferiority; OS, overall survival; PFS, progression-free asurvival; RT, Richter transformation.

Results

Patients, Exposure, and Disposition

- A total of 533 patients were randomized; patient demographics and baseline characteristics were comparable between treatment arms (**Table 1**)
 - Overall, the median age was 66 years, 45% of patients had del(17p) and 64% had del(11q)
 - The median number of prior therapies was 2 in both arms (overall range, 1–12)
- At data cutoff, 46% of acalabrutinib patients and 41% of ibrutinib patients remained on treatment (**Table 2**)

Table 1. Patient Demographics and Baseline Characteristics

Characteristic	Acalabrutinib (n=268)	Ibrutinib n=265
Age, median (range), years	66 (41–89)	65 (28–88)
≥75 years	44 (16.4)	43 (16.2)
Male sex	185 (69.0)	194 (73.2)
ECOG PS score		
0–1	247 (92.2)	243 (91.7)
2	20 (7.5)	22 (8.3)
Bulky disease ≥5 cm	128 (47.8)	136 (51.3)
Rai stage 3 or 4	131 (48.9)	134 (50.6)
Cytogenetic abnormalities		
del(17p)	121 (45.1)	120 (45.3)
del(11q)	167 (62.3)	175 (66.0)
Complex karyotype[a]	124 (46.3)	125 (47.2)
TP53 mutated	100 (37.3)	112 (42.3)
IGHV unmutated	220 (82.1)	237 (89.4)
No. prior therapies, median (range)	2 (1–9)	2 (1–12)
1–3	234 (87.3)	237 (89.4)
≥4	33 (12.3)	28 (10.6)

Data are n (%) unless otherwise specified.
[a]Patients with ≥3 chromosomal abnormalities.
ECOG PS, Eastern Cooperative Oncology Group performance status; IGHV, immunoglobulin heavy chain variable region; *TP53*, tumor protein 53.

Table 2. Patient Disposition

	Acalabrutinib (n=268)	Ibrutinib (n=265)
Duration of follow-up, median (range), months	41.1 (0.0–58.2)	40.7 (0.2–59.1)
Patients who received treatment	265 (98.9)	264 (99.6)[a]
Patients continuing to receive treatment at data cutoff	124 (46.3)	109 (41.1)
Patients who discontinued treatment	141 (52.6)	155 (58.5)
Reasons for treatment discontinuation		
Disease progression[b]	82 (30.6)	68 (25.7)
Adverse event	40 (14.9)	59 (22.3)
Consent withdrawn	7 (2.6)	7 (2.6)
Death	5 (1.9)	6 (2.3)
Investigator decision	5 (1.9)	5 (1.9)
Other	2 (0.7)[c]	10 (3.8)[d]

Data cutoff date: September 15, 2020. Values are reported as n (%) unless stated otherwise.
[a]Includes 1 patient who was randomized to ibrutinib but treated with acalabrutinib and was therefore included in the acalabrutinib arm for safety analyses.
[b]Disease progression includes Richter's transformation.
[c]Includes patients who discontinued treatment due to relocation (n=1) and starting therapy with ibrutinib (n=1) but agreed to remain on study for follow-up.
[d]Includes patients who discontinued treatment due to trial noncompliance (n=2), withdrawal of consent for treatment/follow-up but not considered withdrawal from study per electronic case report guidelines (n=1), refusal of medication (n=1), relocation (n=2), medical monitor decision (n=2), early termination due to second primary malignancy (n=1), and IRC- and medical monitor/sponsor-confirmed progressive disease but investigator disagreed and patient continued ibrutinib off-trial (n=1) but agreed to remain on study for follow-up.

Results (cont'd)

Primary Endpoint

- At a median follow-up of 40.9 months (range, 0.0–59.1), the primary endpoint of non-inferiority on IRC-assessed PFS was met (**Figure 3**)
 - Median PFS was 38.4 months in both arms, and the hazard ratio (HR) was 1.0; the upper bound of the 95% confidence interval of 1.27 was below the non-inferiority criterion of 1.49
- IRC-assessed PFS was generally comparable across prespecified subgroups including patients with del(17p) and del(11q) and regardless of number of prior therapies (**Figure 4**)

Figure 3. IRC-Assessed PFS

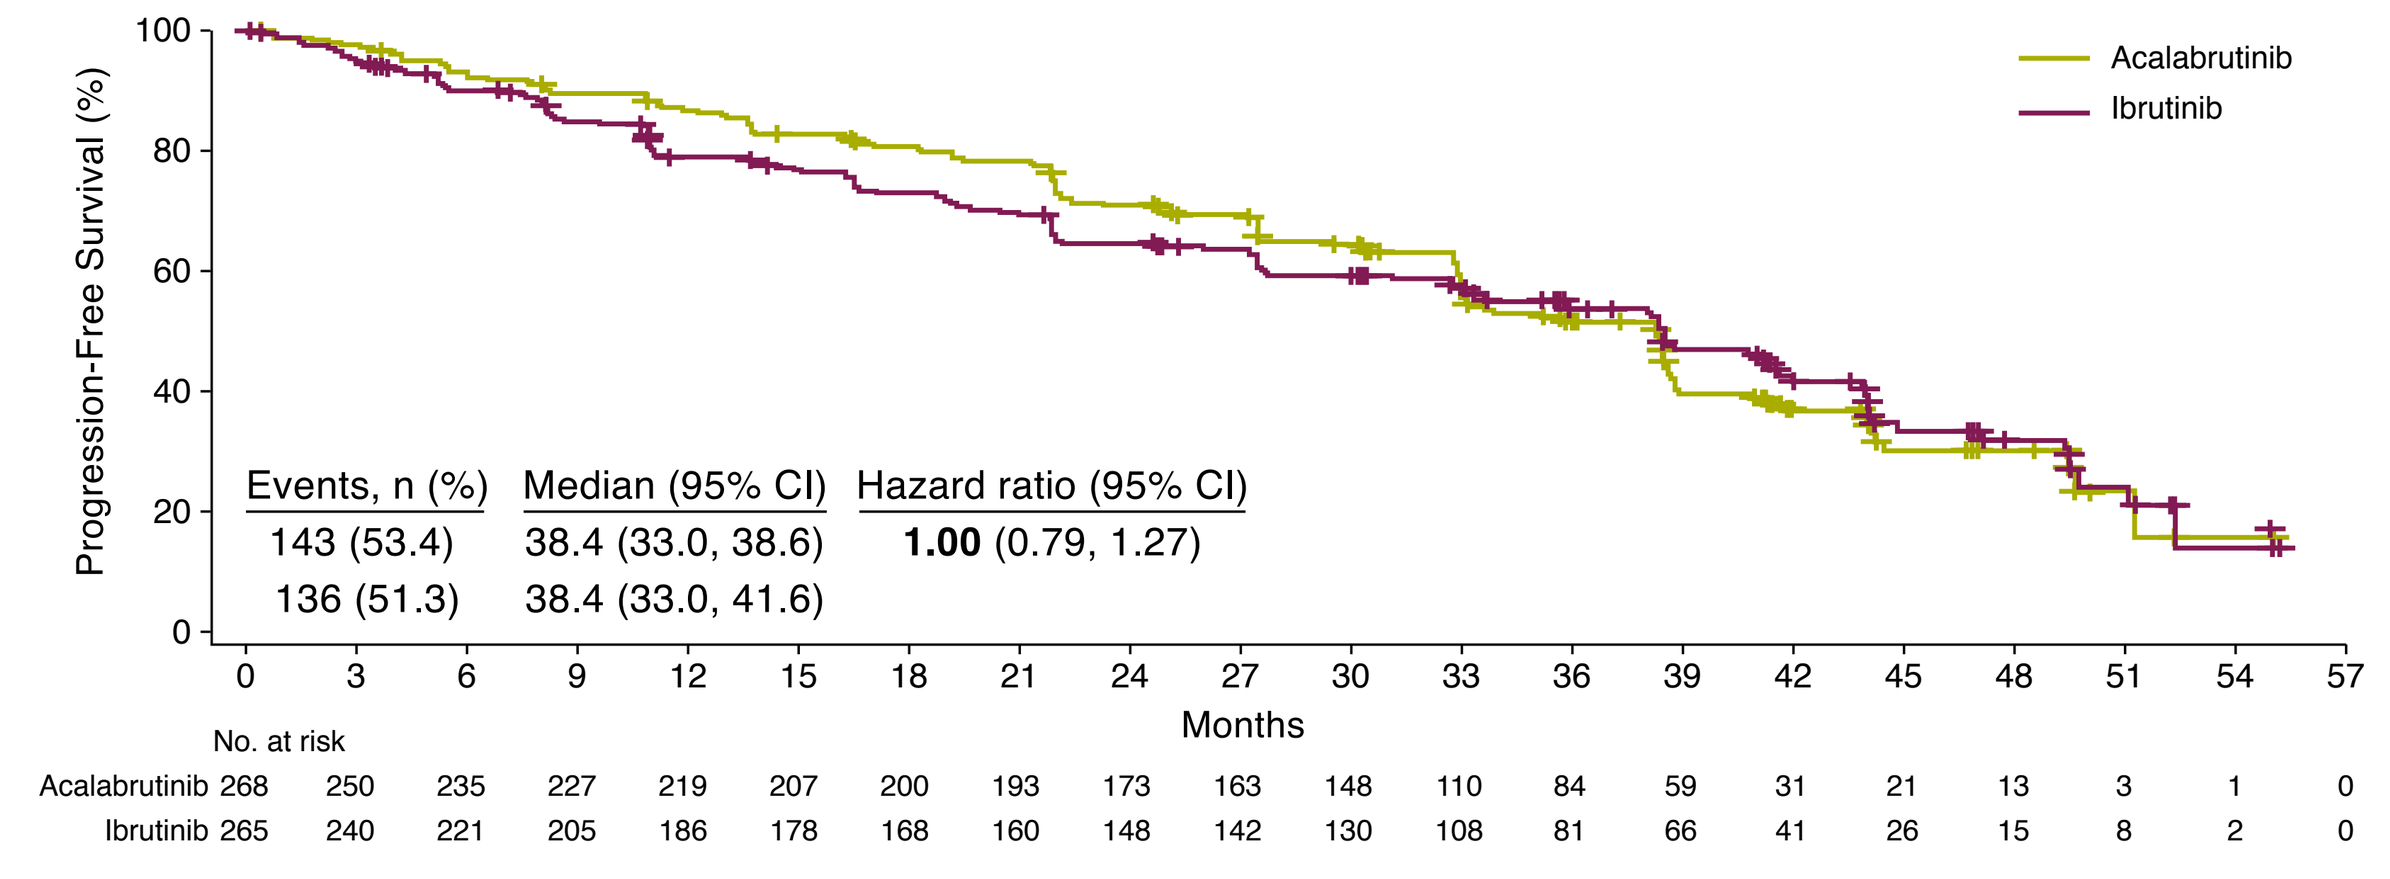
Acalabrutinib
Ibrutinib
Progression-Free Survival (%)
Events, n (%) | Median (95% CI) | Hazard ratio (95% CI)
143 (53.4) | 38.4 (33.0, 38.6) | 1.00 (0.79, 1.27)
136 (51.3) | 38.4 (33.0, 41.6)
Months
No. at risk
Acalabrutinib 268 250 235 227 219 207 200 193 173 163 148 110 84 59 31 21 13 3 1 0
Ibrutinib 265 240 221 205 186 178 168 160 148 142 130 108 81 66 41 26 15 8 2 0

CI, confidence interval; IRC, independent review committee; PFS, progression-free survival.

Figure 4. IRC-Assessed PFS Across Prespecified Subgroups

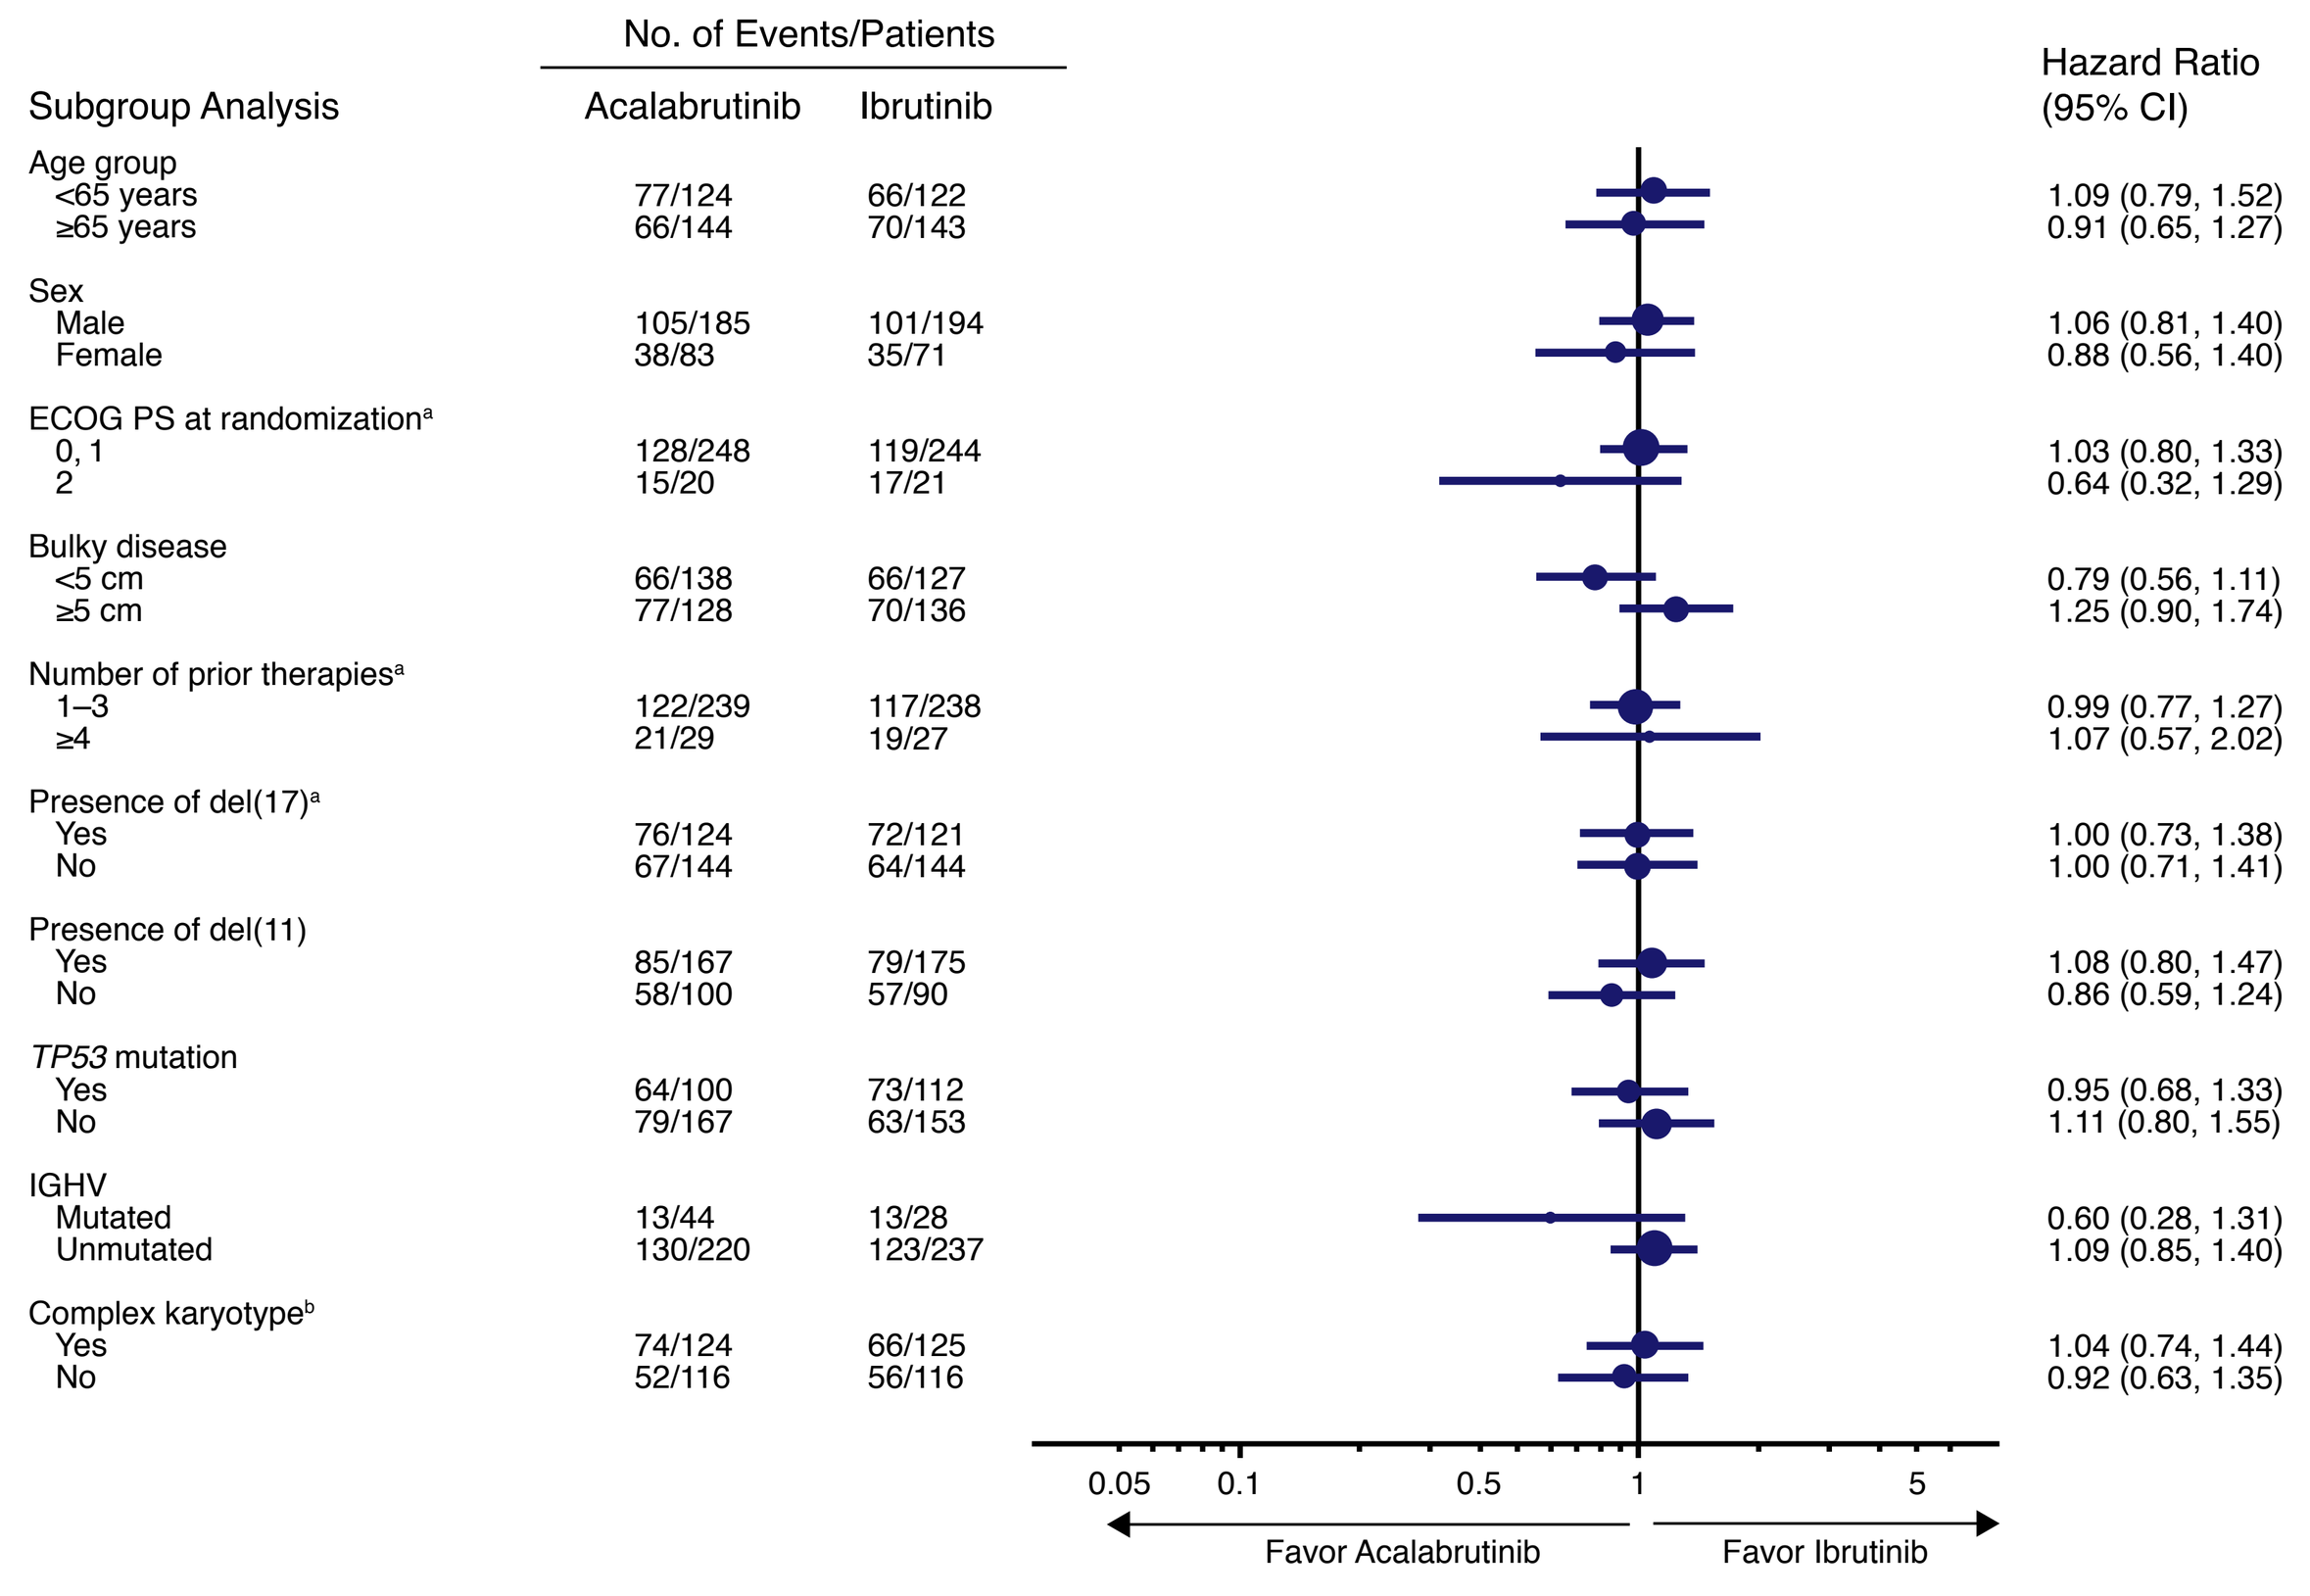
No. of Events/Patients
Subgroup Analysis | Acalabrutinib | Ibrutinib | Hazard Ratio (95% CI)
Age group
<65 years 77/124 66/122 1.09 (0.79, 1.52)
≥65 years 66/144 70/143 0.91 (0.65, 1.27)
Sex
Male 105/185 101/194 1.06 (0.81, 1.40)
Female 38/83 35/71 0.88 (0.56, 1.40)
ECOG PS at randomizationa
0, 1 128/248 119/244 1.03 (0.80, 1.33)
2 15/20 17/21 0.64 (0.32, 1.29)
Bulky disease
<5 cm 66/138 66/127 0.79 (0.56, 1.11)
≥5 cm 77/128 70/136 1.25 (0.90, 1.74)
Number of prior therapiesa
1–3 122/239 117/238 0.99 (0.77, 1.27)
≥4 21/29 19/27 1.07 (0.57, 2.02)
Presence of del(17)a
Yes 76/124 72/121 1.00 (0.73, 1.38)
No 67/144 64/144 1.00 (0.71, 1.41)
Presence of del(11)
Yes 85/167 79/175 1.08 (0.80, 1.47)
No 58/100 57/90 0.86 (0.59, 1.24)
TP53 mutation
Yes 64/100 73/112 0.95 (0.68, 1.33)
No 79/167 63/153 1.11 (0.80, 1.55)
IGHV
Mutated 13/44 13/28 0.60 (0.28, 1.31)
Unmutated 130/220 123/237 1.09 (0.85, 1.40)
Complex karyotypeb
Yes 74/124 66/125 1.04 (0.74, 1.44)
No 52/116 56/116 0.92 (0.63, 1.35)
0.05 0.1 0.5 1 5
Favor Acalabrutinib
Favor Ibrutinib

[a]Randomization stratification factors.
[b]Patients with 3 or more chromosomal abnormalities and one or more structural abnormalities.
CI, confidence interval; ECOG PS, Eastern Cooperative Oncology Group performance status; IRC, independent review committee; PFS, progression-free survival.

Secondary Endpoints

- For the first secondary endpoint tested in the hierarchical testing order, any-grade afib/flutter incidence was significantly lower with acalabrutinib vs ibrutinib (9.4% vs 16.0%; *P*=0.02) (**Table 3**)
 - Median time to any-grade afib/flutter event was longer with acalabrutinib vs ibrutinib
 - Treatment discontinuations due to afib/flutter only occurred in the ibrutinib arm
 - Among patients without a history of afib/flutter, afib/flutter incidence was lower with acalabrutinib vs ibrutinib

Table 3. Atrial Fibrillation/Flutter (Secondary Endpoint)

	Any grade	
	Acalabrutinib (n=266)	Ibrutinib (n=263)
Afib/flutter	**25 (9.4)**[*,a]	**42 (16.0)**[a]
Events/100 person-months	0.366	0.721
Time to onset, median (range), months	28.8 (0.4–52.0)	16.0 (0.5–48.3)
Leading to treatment discontinuation,[b] n/N (%)	0	7/42 (16.7)
Afib/flutter incidence among patients without prior history of afib/flutter, n/N (%)	15/243 (6.2)	37/249 (14.9)

[*]Difference in any-grade incidence rates: −6.6% (95% CI −12.2, −0.9), *P*=0.02.
[a]Grade ≥3 afib/flutter was reported in 13 (4.9%) in the acalabrutinib arm vs 10 (3.8%) in the ibrutinib arm.
[b]Among patients with events of afib/flutter.
Afib/flutter, atrial fibrillation/flutter.

- Incidences of grade ≥3 infections and Richter transformation were comparable for acalabrutinib and ibrutinib
 - Grade ≥3 infections: 31% (n=82) vs 30% (n=79), respectively (*P*=0.8777)
 - Richter transformation: 4% (n=10) vs 5% (n=13)
- Median OS was not reached in either arm (HR, 0.82; 95% CI, 0.59, 1.15; **Figure 5**)

Figure 5. Overall Survival

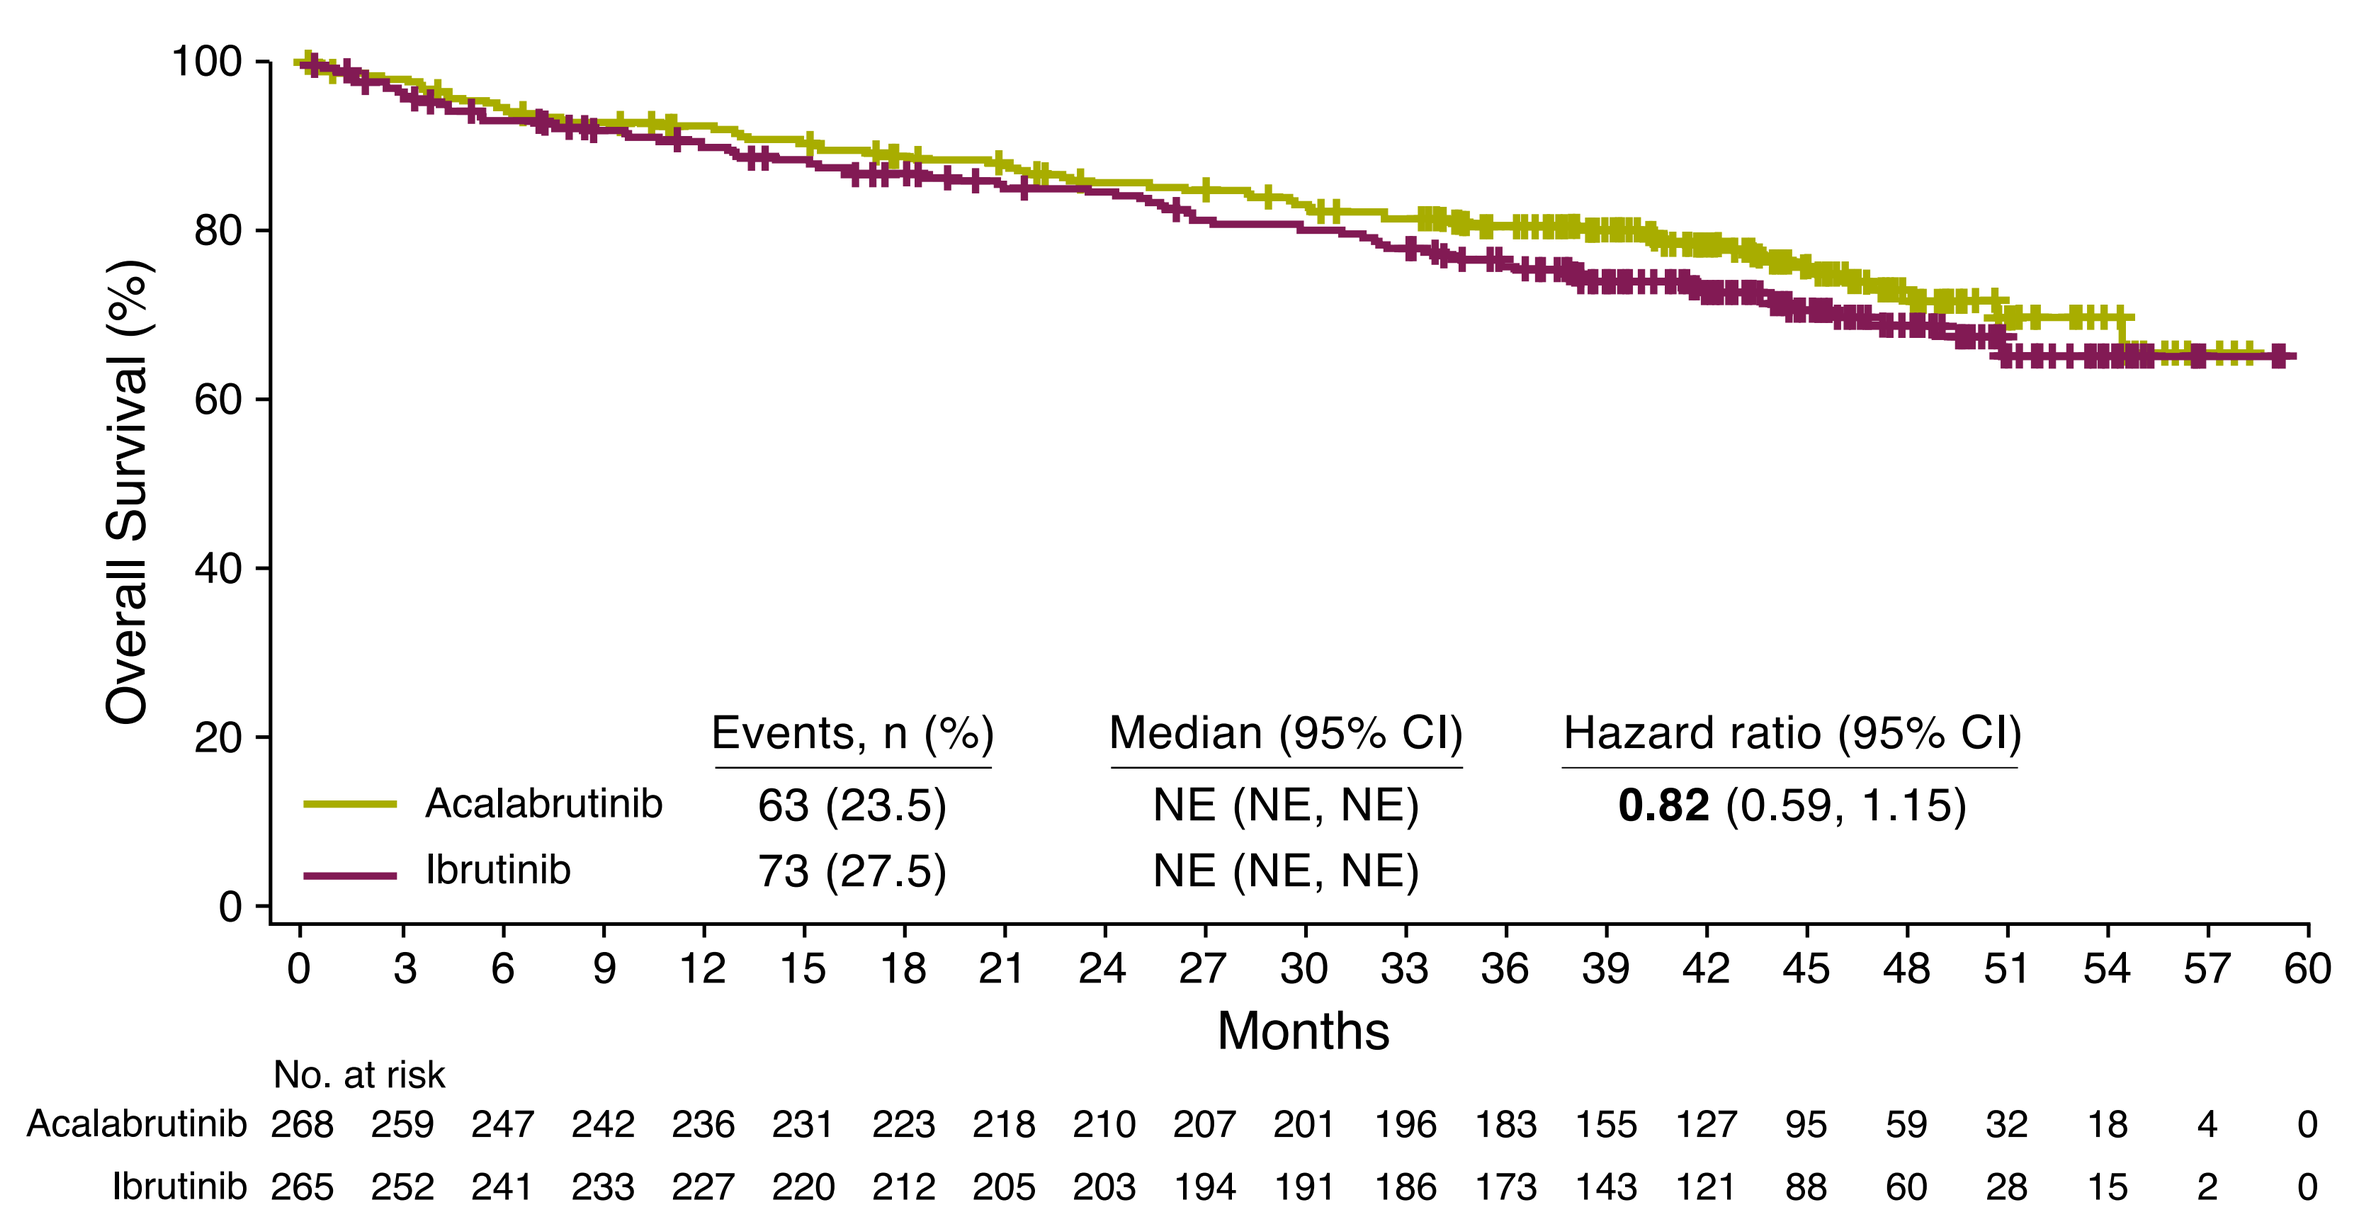
Overall Survival (%)
Events, n (%) | Median (95% CI) | Hazard ratio (95% CI)
Acalabrutinib 63 (23.5) | NE (NE, NE) | 0.82 (0.59, 1.15)
Ibrutinib 73 (27.5) | NE (NE, NE)
Months
No. at risk
Acalabrutinib 268 259 247 242 236 231 223 218 210 207 201 196 183 155 127 95 59 32 18 4 0
Ibrutinib 265 252 241 233 227 220 212 205 203 194 191 186 173 143 121 88 60 28 15 2 0

CI, confidence interval; NE, not estimable.

Safety

- Median treatment exposure duration was slightly longer in the acalabrutinib arm (**Table 4**)
- Incidences of grade ≥3 AEs, AEs leading to treatment discontinuation, serious AEs, and deaths due to AEs were numerically less frequent with acalabrutinib than ibrutinib (**Table 4**)
- Among the most common any-grade AEs in ≥15% of patients in either arm (**Table 5**):
 - Diarrhea, arthralgia, contusion, atrial fibrillation, and hypertension occurred less frequently with acalabrutinib
 - Headache and cough occurred less frequently with ibrutinib

Table 4. Safety Summary

	Any grade	
	Acalabrutinib (n=266)	Ibrutinib (n=263)
Duration of treatment exposure, median (range), months	38.3 (0.3–55.9)	35.5 (0.2–57.7)
Any-grade AEs	260 (97.7)	256 (97.3)
Grade ≥3 AEs	183 (68.8)	197 (74.9)
AEs leading to treatment discontinuation	39 (14.7)	56 (21.3)
Serious AEs	143 (53.8)	154 (58.6)
Deaths due to AEs[a]	17 (6.4)	25 (9.5)

Values are reported as n (%) unless stated otherwise.
[a]Includes deaths occurring within 30 days of last dose; deaths occurring after the start of subsequent anticancer therapy were not included in the assessment of deaths within 30 days of last dose, regardless of time after last dose.
AE, adverse event.

Table 5. Most Common AEs

Events, n (%)	Any grade Acalabrutinib (n=266)	Any grade Ibrutinib (n=263)	Grade ≥3 Acalabrutinib (n=266)	Grade ≥3 Ibrutinib (n=263)
Diarrhea[a,b]	92 (34.6)	**121 (46.0)**	3 (1.1)	**13 (4.9)**
Headache[a,b]	**92 (34.6)**	53 (20.2)	**4 (1.5)**	0
Cough[a]	**77 (28.9)**	56 (21.3)	2 (0.8)	1 (0.4)
URTI	71 (26.7)	65 (24.7)	5 (1.9)	1 (0.4)
Neutropenia	56 (21.1)	65 (24.7)	52 (19.5)	60 (22.8)
Pyrexia	62 (23.3)	50 (19.0)	8 (3.0)	2 (0.8)
Arthralgia[a]	42 (15.8)	**60 (22.8)**	0	2 (0.8)
Hypertension[a,b]	23 (8.6)	**60 (22.8)**	11 (4.1)	**23 (8.7)**
Anemia	58 (21.8)	49 (18.6)	31 (11.7)	34 (12.9)
Fatigue[b]	54 (20.3)	44 (16.7)	**9 (3.4)**	0
Nausea	47 (17.7)	49 (18.6)	0	1 (0.4)
Contusion[a]	31 (11.7)	**48 (18.3)**	0	1 (0.4)
Pneumonia	47 (17.7)	43 (16.3)	28 (10.5)	23 (8.7)
Atrial fibrillation[a]	24 (9.0)	**41 (15.6)**	12 (4.5)	9 (3.4)
Thrombocytopenia	40 (15.0)	35 (13.3)	26 (9.8)	18 (6.8)

Higher incidence in **bold** for terms with statistical differences.
Among most common AEs above, grade 5 were reported in 5 (1.9%) acalabrutinib patients (pyrexia, n=1; pneumonia, n=4) and 4 (1.5%) ibrutinib patients (URTI, n=1; pneumonia, n=3).
[a]Based on Barnard's exact test, two-sided *P*-value <0.05 without multiplicity adjustment for any-grade events.
[b]Based on Barnard's exact test, two-sided *P*-value <0.05 without multiplicity adjustment for grade ≥3 events.
Includes AEs reported at ≥15% incidence (any grade) in either arm.
AE, adverse event; URTI, upper respiratory tract infection.

- Events of clinical interest (ECIs) are reported in **Table 6**
 - ECIs that were statistically different between treatment arms were afib/flutter, overall bleeding events, hypertension, and interstitial lung disease/pneumonitis, which were more frequent in the ibrutinib arm
- Kaplan-Meier (KM) analyses of the cumulative incidences of any-grade afib/flutter and hypertension favored acalabrutinib (HR of 0.52 and 0.34, respectively; **Figure 6**)
- KM analyses of the cumulative incidences of any-grade bleeding events, diarrhea, and arthralgia also favored acalabrutinib (HR of 0.63, 0.61, and 0.61, respectively; **Figure 7**)

Table 6. Events of Clinical Interest

Events, n (%)	Any grade Acalabrutinib (n=266)	Any grade Ibrutinib (n=263)	Grade ≥3 Acalabrutinib (n=266)	Grade ≥3 Ibrutinib (n=263)
Cardiac events	64 (24.1)	79 (30.0)	23 (8.6)	25 (9.5)
Atrial fibrillation[a*]	25 (9.4)	**42 (16.0)**	13 (4.9)	10 (3.8)
Ventricular arrhythmias[b]	0	3 (1.1)	0	1 (0.4)
Bleeding events*	101 (38.0)	**135 (51.3)**	10 (3.8)	12 (4.6)
Major bleeding events[c]	12 (4.5)	14 (5.3)	10 (3.8)	12 (4.6)
Hypertension[d*]	25 (9.4)	**61 (23.2)**	11 (4.1)	**24 (9.1)**
Infections[e]	208 (78.2)	214 (81.4)	82 (30.8)	79 (30.0)
ILD/pneumonitis*	7 (2.6)	**17 (6.5)**	1 (0.4)	2 (0.8)
SPMs excluding NMSC	24 (9.0)	20 (7.6)	16 (6.0)	14 (5.3)

Higher incidence indicated in **bold** for terms with statistical differences.
*Two-sided *P*-value for event comparisons <0.05 without multiplicity adjustment.
[a]Includes events with preferred terms atrial fibrillation and atrial flutter.
[b]Includes events with preferred terms torsade de pointes, ventricular arrhythmia, ventricular extrasystoles, ventricular fibrillation, ventricular flutter, ventricular tachyarrhythmia, and ventricular tachycardia.
[c]Defined as any hemorrhagic event that was serious, grade ≥3 in severity, or a central nervous system hemorrhage (any severity grade).
[d]Included events with the preferred terms of hypertension, blood pressure increased, and blood pressure systolic increased.
[e]Most common grade ≥3 infections were pneumonia (acalabrutinib, 10.5%; ibrutinib, 8.7%), sepsis (1.5% vs 2.7%, respectively), and UTI (1.1% vs 2.3).
ILD, interstitial lung disease; NMSC, nonmelanoma skin cancer; SPMs, second primary malignancies; UTI, urinary tract infection.

Figure 6. Cumulative Incidence of Any-Grade Atrial Fibrillation/Flutter (A) and Hypertension (B)

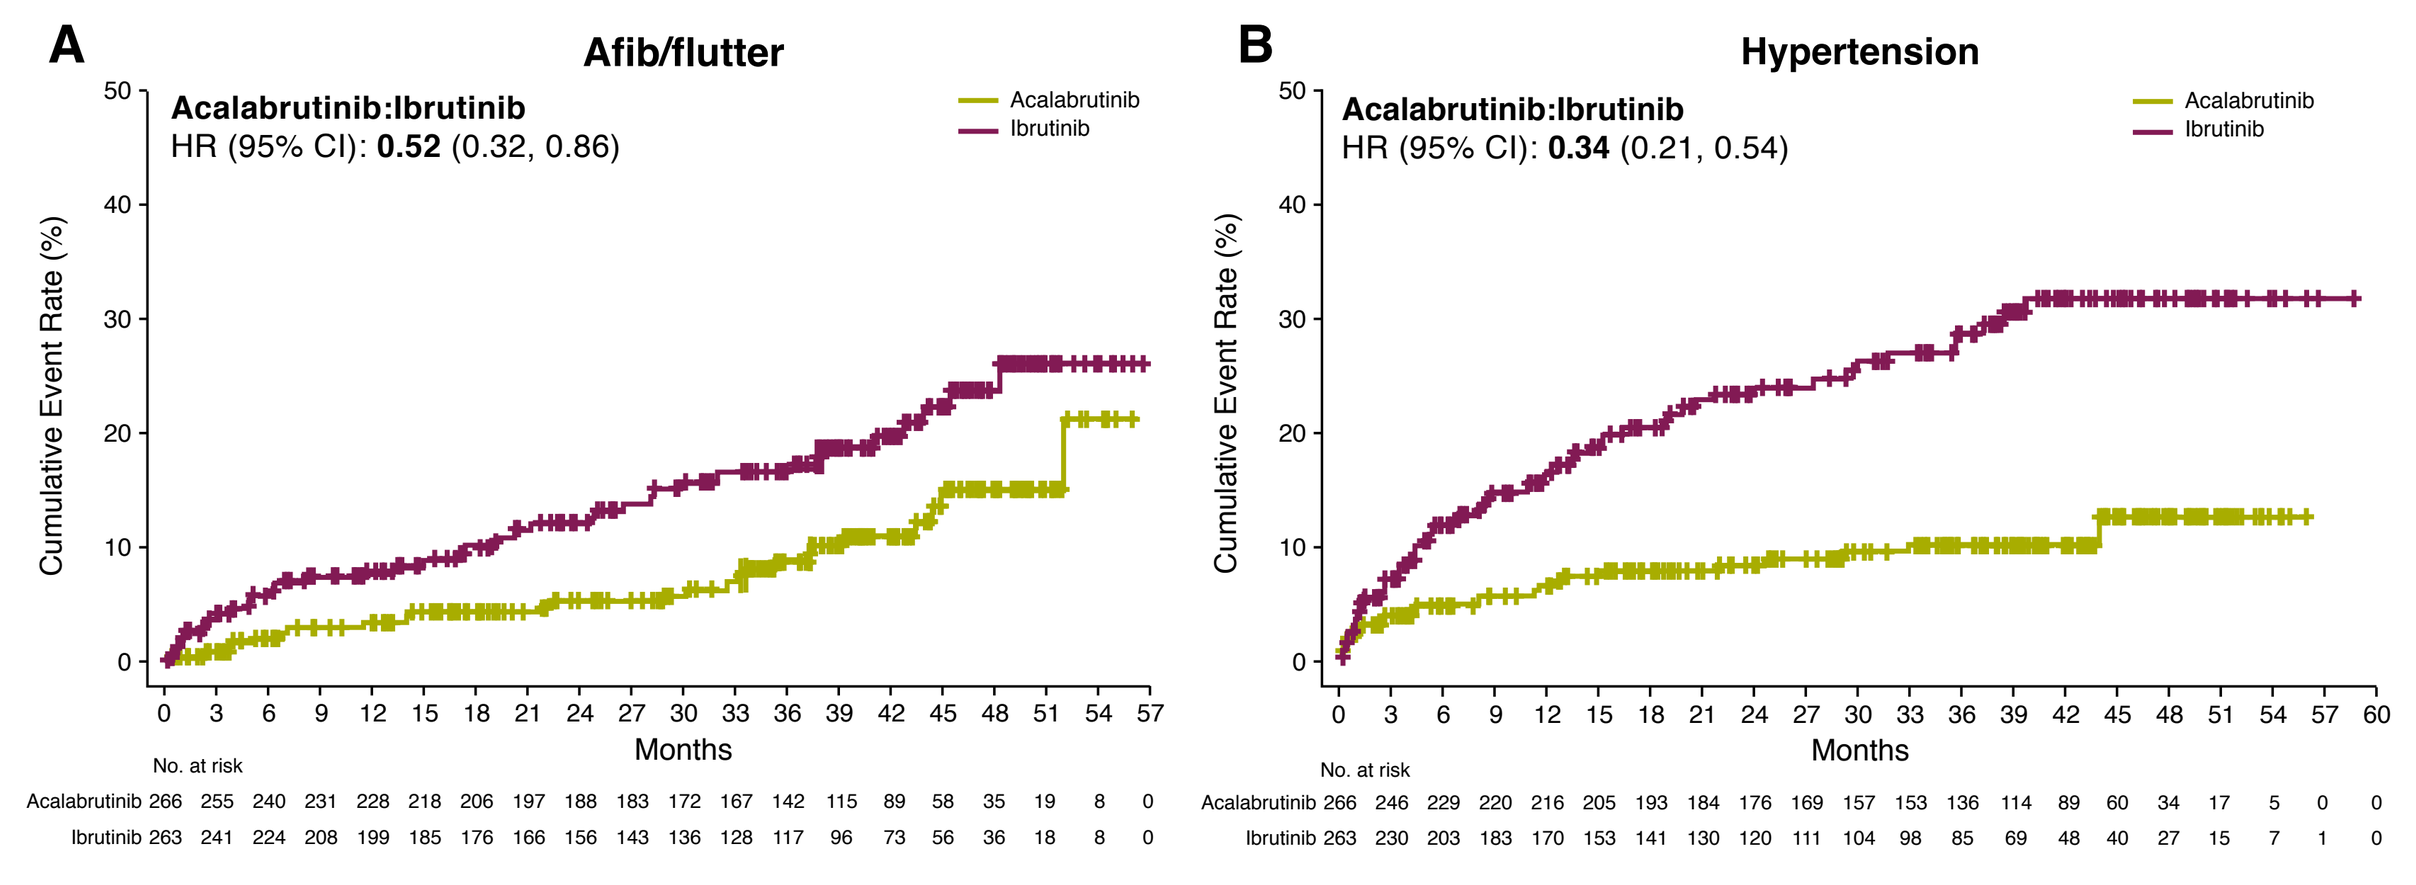
A
Afib/flutter
Acalabrutinib:Ibrutinib
HR (95% CI): 0.52 (0.32, 0.86)
Acalabrutinib
Ibrutinib
Cumulative Event Rate (%)
Months
No. at risk
Acalabrutinib 266 255 240 231 228 218 206 197 188 183 172 167 142 115 89 58 35 19 8 0
Ibrutinib 263 241 224 208 199 185 176 166 156 143 136 128 117 96 73 56 36 18 8 0
B
Hypertension
Acalabrutinib:Ibrutinib
HR (95% CI): 0.34 (0.21, 0.54)
Acalabrutinib
Ibrutinib
Cumulative Event Rate (%)
Months
No. at risk
Acalabrutinib 266 246 229 220 216 205 193 184 176 169 157 153 136 114 89 60 34 17 5 0 0
Ibrutinib 263 230 203 183 170 153 141 130 120 111 104 98 85 69 48 40 27 15 7 1 0

CI, confidence interval; HR, hazard ratio.

Figure 7. Cumulative Incidence of Any-Grade Bleeding (A), Diarrhea (B), and Arthralgia (C)

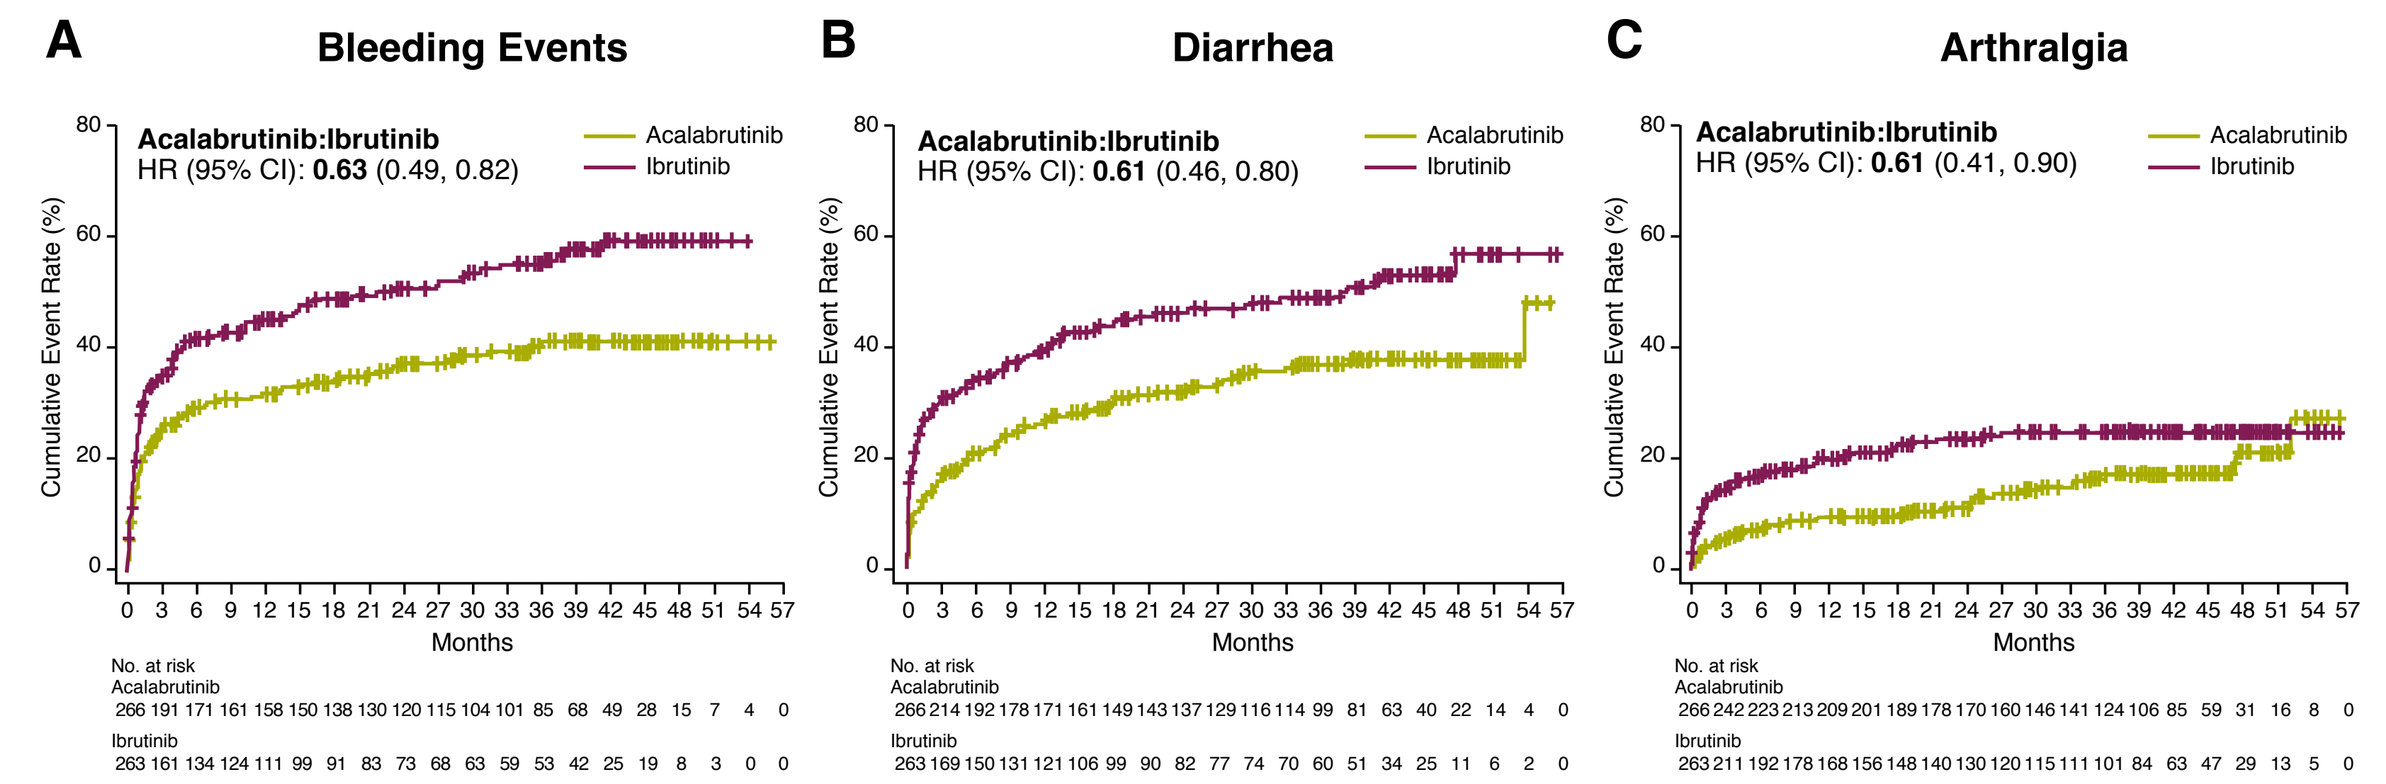
A
Bleeding Events
Acalabrutinib:Ibrutinib
HR (95% CI): 0.63 (0.49, 0.82)
B
Diarrhea
Acalabrutinib:Ibrutinib
HR (95% CI): 0.61 (0.46, 0.80)
C
Arthralgia
Acalabrutinib:Ibrutinib
HR (95% CI): 0.61 (0.41, 0.90)
Acalabrutinib
Ibrutinib
Cumulative Event Rate (%)
Months
No. at risk

CI, confidence interval; HR, hazard ratio.

References

1. Vitale C, Burger JA. *Expert Opin Pharmacother.* 2016;17:1077-89. **2.** Lougaris V, et al. *J Allergy Clin Immunol.* 2014;133:1644-50. **3.** de Gorter DJ, et al. *Immunity.* 2007;26:93-104. **4.** Barf T, et al. *J Pharmacol Exp Ther.* 2017;363:240-52. **5.** Imbruvica [package insert]. Pharmacyclics, Janssen Biotech, Inc.; 2020. **6.** Byrd JC, et al. *Blood.* 2019;133:2031-42. **7.** Mato AR, et al. *Haematologica.* 2018;103:874-9. **8.** Dickerson T, et al. *Blood.* 2019;134:1919-28. **9.** Byrd JC, et al. *N Engl J Med.* 2016;374:323-32. **10.** Bond DA, Woyach JA. *Curr Hematol Malig Rep.* 2019;14:197-205. **11.** Caldeira D, et al. *PLoS One.* 2019;14:e0211228. **12.** Caron F, et al. *Blood Adv.* 2017;1:772-8. **13.** Calquence [package insert]. AstraZeneca Pharmaceuticals, 2019. **14.** Hallek M, et al. *Blood.* 2008;111:5446-56.

Acknowledgements

We thank all patients who participated in this study, their families, and the investigators, from 124 centers in 15 countries. The authors also thank the members of the Acerta Pharma study team who designed this trial, including Raquel Izumi, PhD, Ahmed Hamdy, MD, and Wayne Rothbaum. This study was sponsored by Acerta Pharma, a member of the AstraZeneca Group. Medical writing support was provided by Peloton Advantage, LLC (Parsippany, NJ, USA), an OPEN Health company and funded by Acerta Pharma, a member of the AstraZeneca Group. Figures 3, 4, 5 and 6 borrowed with permission from Byrd JC, et al. Acalabrutinib versus ibrutinib in previously treated chronic lymphocytic leukemia: results of the first randomized phase III trial. *J Clin Oncol.* 2021. Online ahead of print. https://ascopubs.org/doi/full/10.1200/JCO.21.01210. ©American Society of Clinical Oncology.

Disclosures

JC Byrd: Stock and Other Ownership Interests: Vincerx Pharma. Honoraria: Pharmacyclics, AstraZeneca, Novartis, Syndax, Trillium Therapeutics. Consulting or Advisory Role: Acerta Pharma, Janssen, Kura Oncology, Novartis, Syndax, AstraZeneca. Research Funding: Acerta Pharma, Pharmacyclics, Zencor. Patents, Royalties, Other Intellectual Property: OSU Patents. Travel, Accommodations, Expenses: Gilead Sciences, Janssen, Novartis, Pharmacyclics, TG Therapeutics. **P Hillmen:** Honoraria: Janssen, AbbVie, Roche. Research Funding: Janssen, Pharmacyclics, Roche, Gilead Sciences, AbbVie. Travel, Accommodations, Expenses: Janssen, AbbVie. **P Ghia:** Honoraria: AbbVie, BeiGene, Janssen Oncology, Gilead Sciences, Juno Therapeutics, Sunesis Pharmaceuticals, ArQule, Adaptive Biotechnologies, Dynamo Therapeutics, MEI Pharma, Acerta Pharma/AstraZeneca, Juno/ Celgene/Bristol Myers Squibb, MSD, Lilly, Roche. Consulting or Advisory Role: AbbVie, BeiGene, Janssen, Gilead Sciences, Sunesis Pharmaceuticals, Juno Therapeutics, ArQule, Adaptive Biotechnologies, Dynamo Therapeutics, MEI Pharma, Acerta Pharma/AstraZeneca, MSD, Lilly, Roche. Research Funding: AbbVie, Janssen Oncology, Gilead Sciences, Sunesis Pharmaceuticals, Novartis, AstraZeneca. **AP Kater:** Consulting or Advisory Role: Janssen Oncology, AbbVie/Genentech, AstraZeneca, Bristol Myers Squibb/Celgene, Lava Therapeutics. Research Funding: Janssen Oncology, Roche/Genentech, Bristol Myers Squibb/ Celgene, AbbVie, AstraZeneca. Travel, Accommodations, Expenses: Acerta Pharma/AstraZeneca, Roche/ Genentech. **A Chanan-Khan:** Stock and Other Ownership Interests: Matthew & Asher Inc, NanoDev Therapeutics, STARTON Therapeutics. Honoraria: BeiGene, Ascentage Pharma. Research Funding: Ascentage Pharma. Patents, Royalties, Other Intellectual Property: Patent on PSMB9 biomarker. **RR Furman:** Honoraria: Janssen, AstraZeneca. Consulting or Advisory Role: Pharmacyclics, Janssen Biotech, Genentech/ Roche, Loxo, TG Therapeutics, Verastem, Acerta Pharma, AstraZeneca, Beigene, Incyte, OncoTracker, AbbVie, MorphoSys, Sanofi. Research Funding: Acerta Pharma, TG Therapeutics. Expert Testimony: AbbVie, Janssen Oncology. Travel, Accommodations, Expenses: TG Therapeutics, Janssen Oncology. Other Relationship: Incyte, Janssen Biotech. **S O'Brien:** Employment: University of California, Irvine. Honoraria: Celgene, Janssen, Pharmacyclics, Gilead Sciences, Pfizer, Amgen, Astellas Pharma, GlaxoSmithKline, Aptose Biosciences, Vaniam Group, AbbVie, Sunesis Pharmaceuticals, Alexion Pharmaceuticals, Eisai, TG Therapeutics, Nova Research Company. Consulting or Advisory Role: Amgen, Celgene, GlaxoSmithKline, Janssen Oncology, Aptose Biosciences, Vaniam Group, AbbVie/Genentech, Sunesis Pharmaceuticals, Alexion Pharmaceuticals, Astellas Pharma, Gilead Sciences, Pharmacyclics, TG Therapeutics, Pfizer, Sunesis Pharmaceuticals. Research Funding: Acerta Pharma, Regeneron, Gilead Sciences, Pfizer, TG Therapeutics, Pfizer, Kite, a Gilead company, Sunesis Pharmaceuticals Travel, Accommodations, Expenses: Celgene, Janssen, Gilead Sciences, Regeneron, Janssen Oncology. **MN Yenerel:** Consulting or Advisory Role: Pfizer, Alexion Pharmaceuticals. Speakers' Bureau: Janssen Oncology. Travel, Accommodations, Expenses: Pfizer. **A Illés:** Consulting or Advisory Role: Janssen, Takeda, Novartis, Pfizer, Roche, Celgene. Research Funding: Takeda, Seattle Genetics. Travel, Accommodations, Expenses: Novartis, Janssen, Pfizer, Roche. **N Kay:** Consulting or Advisory Role: MorphoSys, Celgene, Agios, CytomX Therapeutics, AstraZeneca, Pharmacyclics, DAVA Pharmaceuticals, Juno Therapeutics, Rigel, Oncotracker, Bristol Myers Squibb, AbbVie, Targeted Oncology, Acerta Pharma/AstraZeneca, MEI Pharma, Sunesis Pharmaceuticals, TG Therapeutics, Tolero Pharmaceuticals, Janssen Biotech, Genentech/AbbVie. Research Funding: Pharmacyclics/Janssen, Tolero Pharmaceuticals, Acerta Pharma, MEI Pharma, Celgene, Genentech, Sunesis Pharmaceuticals, AbbVie, TG Therapeutics, Bristol Myers Squibb. **JA Garcia-Marco:** Consulting or Advisory Role: AbbVie, Janssen, AstraZeneca Spain. Speakers' Bureau: AbbVie, Janssen, AstraZeneca Spain. Research Funding: AbbVie, Janssen. Travel, Accommodations, Expenses: AbbVie, Janssen. **A Mato:** Consulting or Advisory Role: TG Therapeutics, AbbVie/Genentech, Celgene, Pharmacyclics, Adaptive Biotechnologies, Verastem, Johnson & Johnson, Acerta Pharma/AstraZeneca, DTRM, Loxo/Lilly, Curio/Vaniam Group, Merck, Bristol Myers Squibb/Pfizer. Research Funding: Regeneron, TG Therapeutics, Sunesis Pharmaceuticals, LOXO, AbbVie/Genentech, Pharmacyclics, Adaptive Biotechnologies, Johnson & Johnson, Acerta Pharma/AstraZeneca, DTRM, Genmab, Nurix. **J Pinilla-Ibarz:** Honoraria: Novartis, Pharmacyclics, Janssen, AbbVie, Takeda, AstraZeneca Consulting or Advisory Role: Novartis, Pharmacyclics, Janssen, AbbVie Speakers' Bureau: Pharmacyclics/Janssen, AbbVie, Takeda, AstraZeneca Patents, Royalties, Other Intellectual Property: From a WT vaccine patent by Memorial Sloan Kettering Cancer Center. **JF Seymour:** Honoraria: AbbVie, Acerta Pharma, Janssen, Roche, Sunesis Pharmaceuticals, Takeda. Consulting or Advisory Role: AbbVie, Acerta Pharma, Janssen, Roche, Sunesis Pharmaceuticals, Takeda, AstraZeneca, BMS, Gilead Sciences, MEI Pharma, MorphoSys. Speakers' Bureau: AbbVie, Roche. Research Funding: AbbVie, Celgene, Janssen, Roche. Expert Testimony: Roche. Travel, Accommodations, Expenses: AbbVie, Roche. **S Lepretre** has nothing to disclose. **S Stilgenbauer:** Honoraria: AbbVie, AstraZeneca, Celgene, Gilead Sciences, GlaxoSmithKline, Roche, Janssen. Consulting or Advisory Role: AbbVie, AstraZeneca, Celgene, Gilead Sciences, GlaxoSmithKline, Roche, Janssen. Speakers' Bureau: AbbVie, AstraZeneca, Celgene, Gilead Sciences, GlaxoSmithKline, Roche, Janssen. Research Funding: AbbVie, AstraZeneca, Celgene, Gilead Sciences, GlaxoSmithKline, Roche, Janssen. Travel, Accommodations, Expenses: AbbVie, AstraZeneca, Celgene, Gilead Sciences, GlaxoSmithKline, Roche, Janssen. **T Robak:** Honoraria: AbbVie. Consulting or Advisory Role: AbbVie. Research Funding: AbbVie/Genentech. **P Patel:** Employment: AstraZeneca, Neoleukin Therapeutics. Leadership: Neoleukin Therapeutics. Stock and Other Ownership Interests: AstraZeneca, Neoleukin Therapeutics. **K Higgins:** Employment: AstraZeneca, PROMETRIKA LLC. **S Sohoni:** Employment: AstraZeneca, Portola Pharmaceuticals. Stock and Other Ownership Interests: Theravance. **W Jurczak:** Consulting or Advisory Role: Janssen-Cilag, Roche, AstraZeneca, Debiopharm Group, Epizyme. Research Funding: Acerta Pharma, TG Therapeutics, Incyte, Bayer, Sandoz- Novartis, Roche, Takeda, Epizyme, Janssen-Cilag, BeiGene, Debiopharm Group, MorphoSys, MEI Pharma.