Poster #CLL-045

Long-Term Follow-up, Up to 7 Years, in the RESONATE-2 Study of First-Line Ibrutinib Treatment for Chronic Lymphocytic Leukemia (CLL)

Jan Burger, MD, PhD[1]; Paul Barr, MD[2]; Carolyn Owen, MD[3]; Tadeusz Robak, MD, PhD[4]; Alessandra Tedeschi, MD[5]; Osnat Bairey, MD[6]; Peter Hillmen, MBChB, PhD[7]; Steve Coutre, MD[8]; Stephen Devereux, FRCP, FRCPath, PhD, MD[9]; Sebastian Grosicki, MD, PhD[10]; Helen McCarthy, MBBS, PhD, MRCP, FRCPath[11]; Jianyong Li, MD, PhD[12]; Fritz Offner, MD, PhD[13]; Carol Moreno, MD, PhD[14]; Sandra Dai, PhD, MS[15]; Anita Szoke, MD[15]; James P. Dean, MD, PhD[15]; Thomas J. Kipps, MD, PhD[16]; Paolo Ghia, MD, PhD[17]

[1]Department of Leukemia, University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Wilmot Cancer Institute, University of Rochester Medical Center, Rochester, NY, USA; [3]Tom Baker Cancer Centre, University of Calgary, Calgary, AB, Canada; [4]Medical University of Lodz, Copernicus Memorial Hospital, Lodz, Poland; [5]ASST Grande Ospedale Metropolitano Niguarda, Milan, Italy; [6]Rabin Medical Center, Petah Tikva, Israel; [7]The Leeds Teaching Hospitals, St. James Institute of Oncology, Leeds, UK; [8]Stanford Cancer Center, Stanford University School of Medicine, Stanford, CA, USA; [9]Kings College Hospital, NHS Foundation Trust, London, UK; [10]School of Public Health, Silesian Medical University, Katowice, Poland; [11]Royal Bournemouth General Hospital, Bournemouth, UK; [12]Jiangsu Province Hospital, Nanjing, China; [13]Universitair Ziekenhuis Gent, Gent, Belgium; [14]Hospital de la Santa Creu i Sant Pau, Autonomous University of Barcelona, Barcelona, Spain; [15]Pharmacyclics LLC, an AbbVie Company, Sunnyvale, CA, USA; [16]UCSD Moores Cancer Center, San Diego, CA, USA; [17]Università Vita-Salute San Raffaele and IRCCS Ospedale San Raffaele, Milan, Italy

INTRODUCTION

- Ibrutinib, a once-daily Bruton's tyrosine kinase (BTK) inhibitor, is approved in the US for the treatment of patients with chronic lymphocytic leukemia/small lymphocytic lymphoma (CLL/SLL).[1]
 - Ibrutinib is the only therapy for CLL/SLL to demonstrate both a significant progression-free survival (PFS)[2-7] and overall survival (OS)[2,3,6,7] benefit in multiple randomized phase 3 studies for patients with first-line or relapsed/refractory CLL/SLL.
 - Real-world ibrutinib data in CLL are generally consistent with clinical trial results.[8-20]
- At last update, median PFS for ibrutinib was not reached; 70% of patients with CLL treated with first-line ibrutinib remain progression-free and alive.[21]
- Given the durable efficacy observed with single-agent ibrutinib treatment, ongoing evaluations of long-term efficacy and safety are critical to inform clinical decision-making.

OBJECTIVE

- To describe the long-term efficacy and safety with up to 7 years of follow-up from RESONATE-2, the first randomized phase 3 study to establish efficacy of first-line ibrutinib treatment in CLL/SLL

METHODS

- RESONATE-2 is a phase 3, open-label, multicenter, international, randomized study (PCYC-1115/1116; NCT01722487) in first-line CLL/SLL (**Figure 1**).[2, 21]
- Long-term efficacy (PFS, overall response rate [ORR]) is reported per the investigator.
 - After a median follow-up of 5 years, the protocol was amended to provide up to 10 years of PFS follow-up for both arms, and up to 10 years of OS follow-up for the ibrutinib arm only.
- Safety data are reported for ongoing ibrutinib treatment arm only.

Figure 1. RESONATE-2 Study Schema

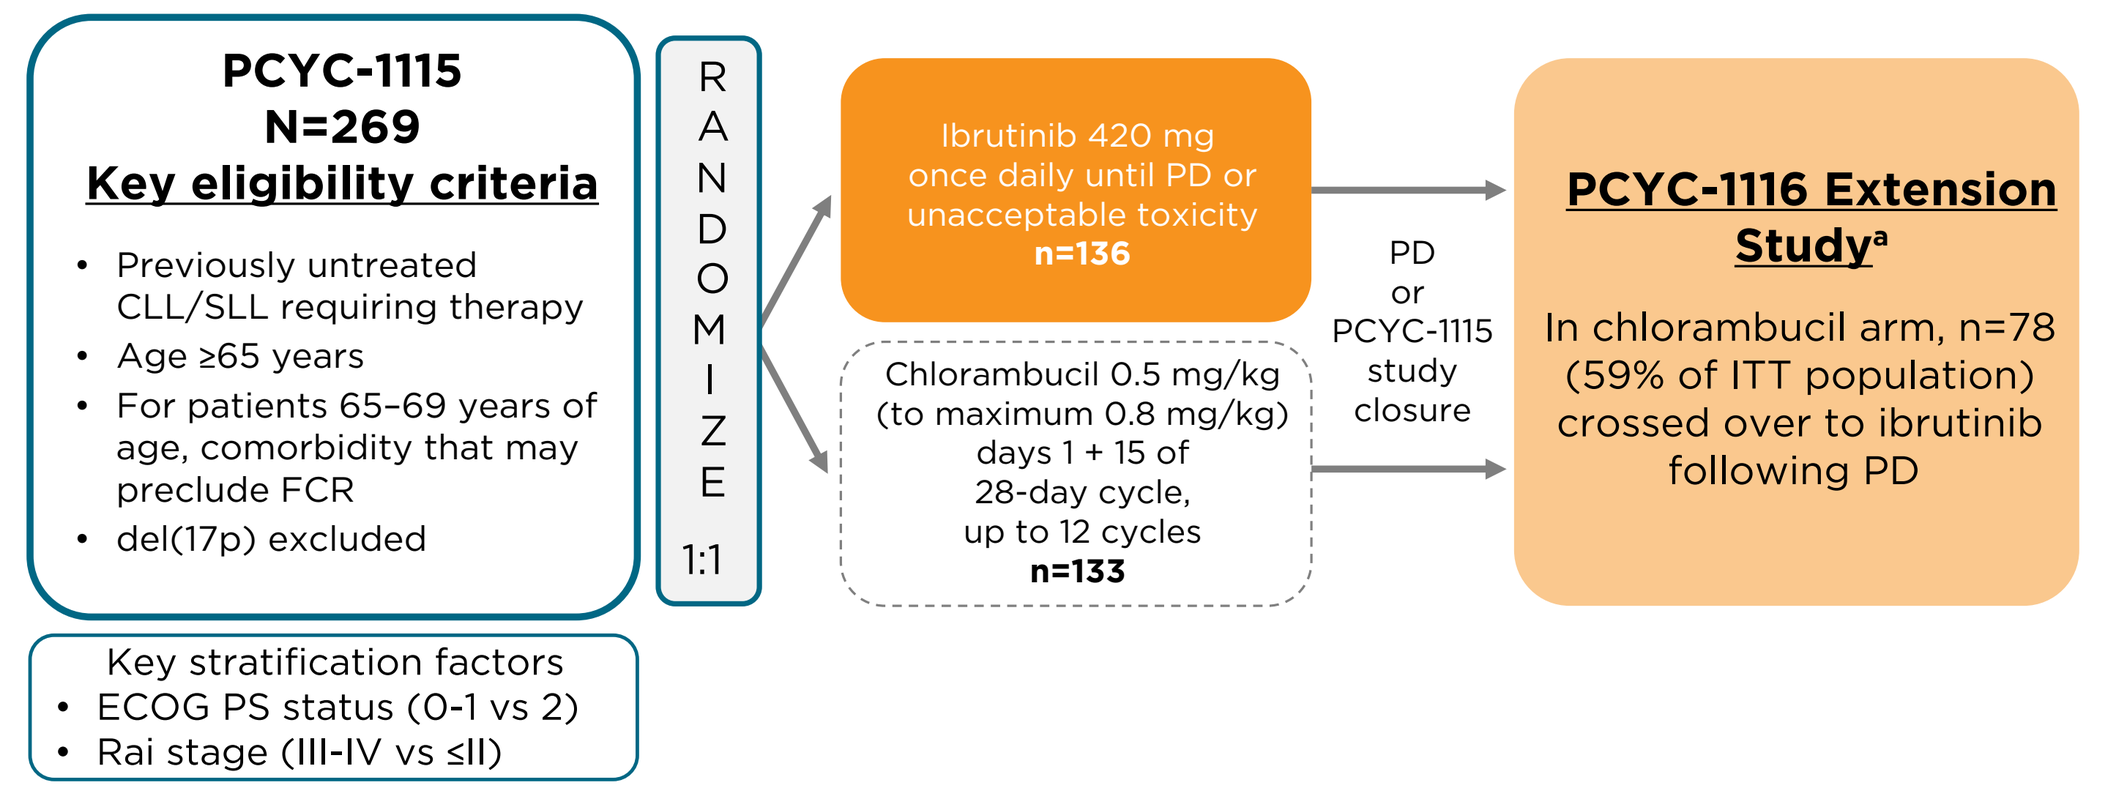
PCYC-1115
N=269
Key eligibility criteria
Previously untreated CLL/SLL requiring therapy
Age ≥65 years
For patients 65–69 years of age, comorbidity that may preclude FCR
del(17p) excluded
Key stratification factors
ECOG PS status (0-1 vs 2)
Rai stage (III-IV vs ≤II)
RANDOMIZE
1:1
Ibrutinib 420 mg once daily until PD or unacceptable toxicity
n=136
Chlorambucil 0.5 mg/kg (to maximum 0.8 mg/kg) days 1 + 15 of 28-day cycle, up to 12 cycles
n=133
PD or PCYC-1115 study closure
PCYC-1116 Extension Study[a]
In chlorambucil arm, n=78 (59% of ITT population) crossed over to ibrutinib following PD

[a]Patients could enroll in separate extension study PCYC-1116 after IRC-confirmed PD or at study PCYC-1115 closure for continuing treatment and follow-up.
ECOG PS, Eastern Cooperative Oncology Group performance status; FCR, fludarabine + cyclophosphamide + rituximab; IRC, independent review committee; ITT, intention-to treat, PD, disease progression.

RESULTS

- Baseline patient characteristics were well-matched and the majority of patients were aged 70 years or older (median 73 years), 35% had bulky disease, 39% had anemia (Hb <11 g/dL), and 65% had β_2-microglobulin (B2M) >3.5 mg/L.
- 22% (54/251) of evaluable patients had del(11q), 58% (118/204) had unmutated IGHV.
- 53% (142/269) of all patients in the ITT population had one or more high-risk features (*TP53* mutation, del[11q], and/or unmutated IGHV).

RESULTS (CONT.)

Efficacy

Figure 2. PFS: Ibrutinib vs Chlorambucil

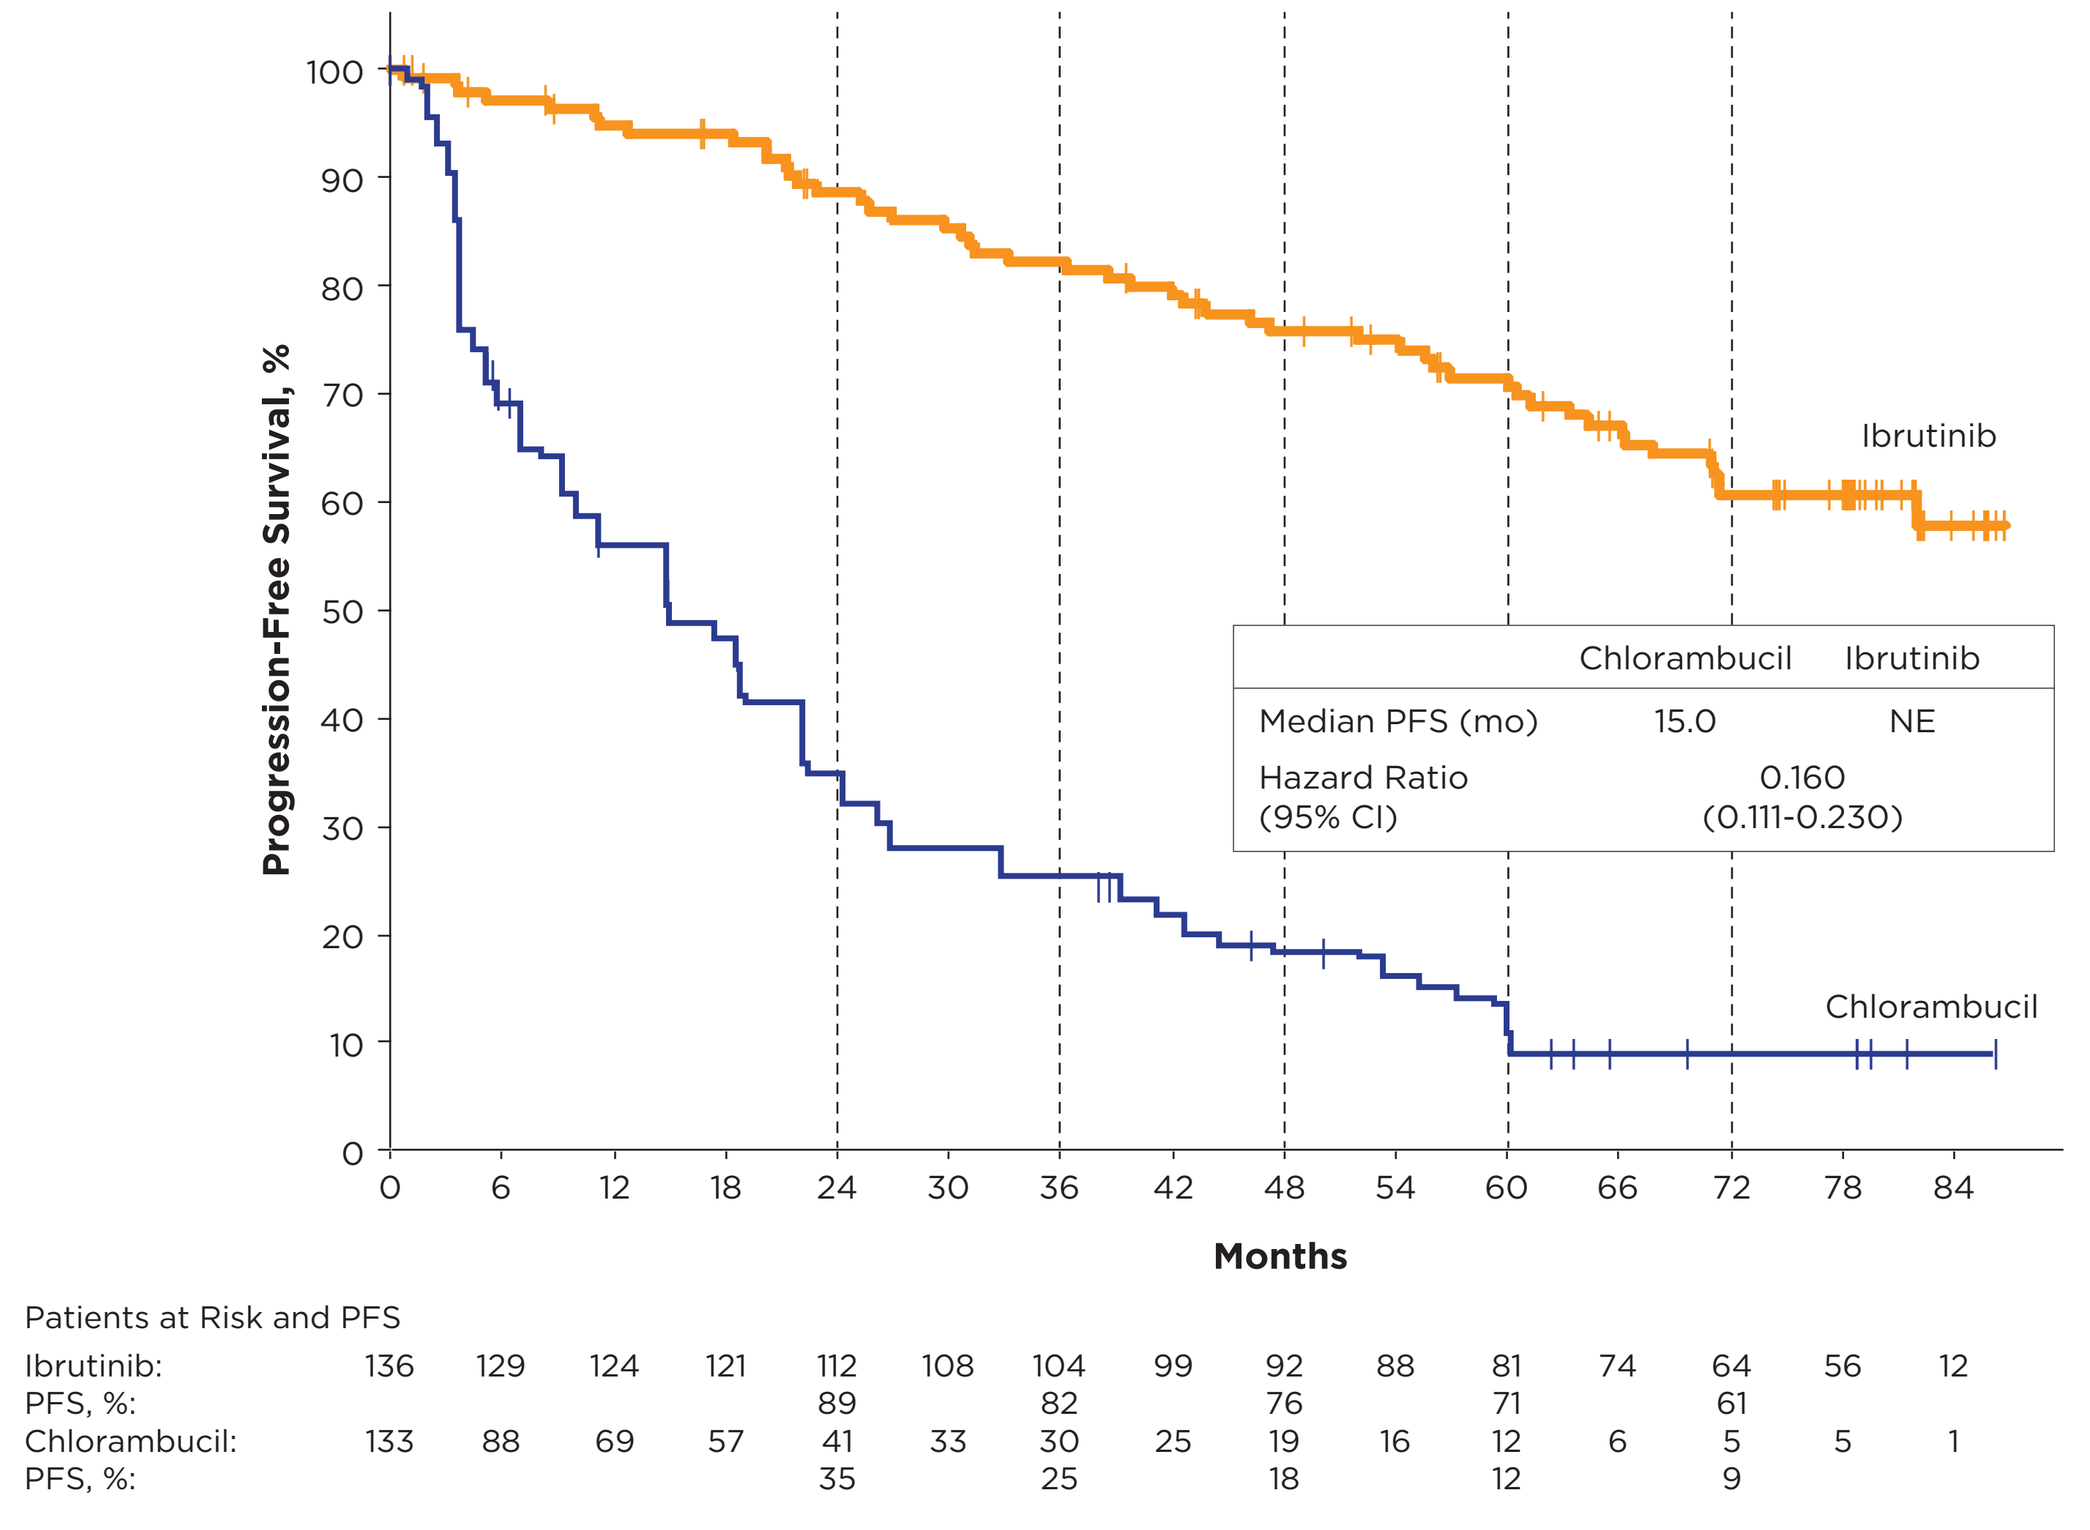
Progression-Free Survival, %
Ibrutinib
Chlorambucil
Chlorambucil Ibrutinib
Median PFS (mo) 15.0 NE
Hazard Ratio (95% CI) 0.160 (0.111-0.230)
Months
Patients at Risk and PFS
Ibrutinib: 136 129 124 121 112 108 104 99 92 88 81 74 64 56 12
PFS, %: 89 82 76 71 61
Chlorambucil: 133 88 69 57 41 33 30 25 19 16 12 6 5 5 1
PFS, %: 35 25 18 12 9

HR, hazard ratio.
HR from stratified Cox regression model.

- With up to 7 years of follow-up, the median PFS was not reached for the ibrutinib arm (**Figure 2**).
 - Ibrutinib-treated patients had an 84% reduction in risk of progression or death.
- At 6.5 years, 61% vs 9% of ibrutinib vs chlorambucil patients were estimated to be progression free and alive.
 - Only 12% of patients randomized to ibrutinib progressed while on active ibrutinib treatment.
- Superior benefit for ibrutinib vs chlorambucil was maintained across subgroup analysis factors, including high-risk genomic features and other baseline clinical characteristics such as advanced stage and bulky disease (**Figure 3**).

Figure 3. PFS in Patient Subgroups of Interest

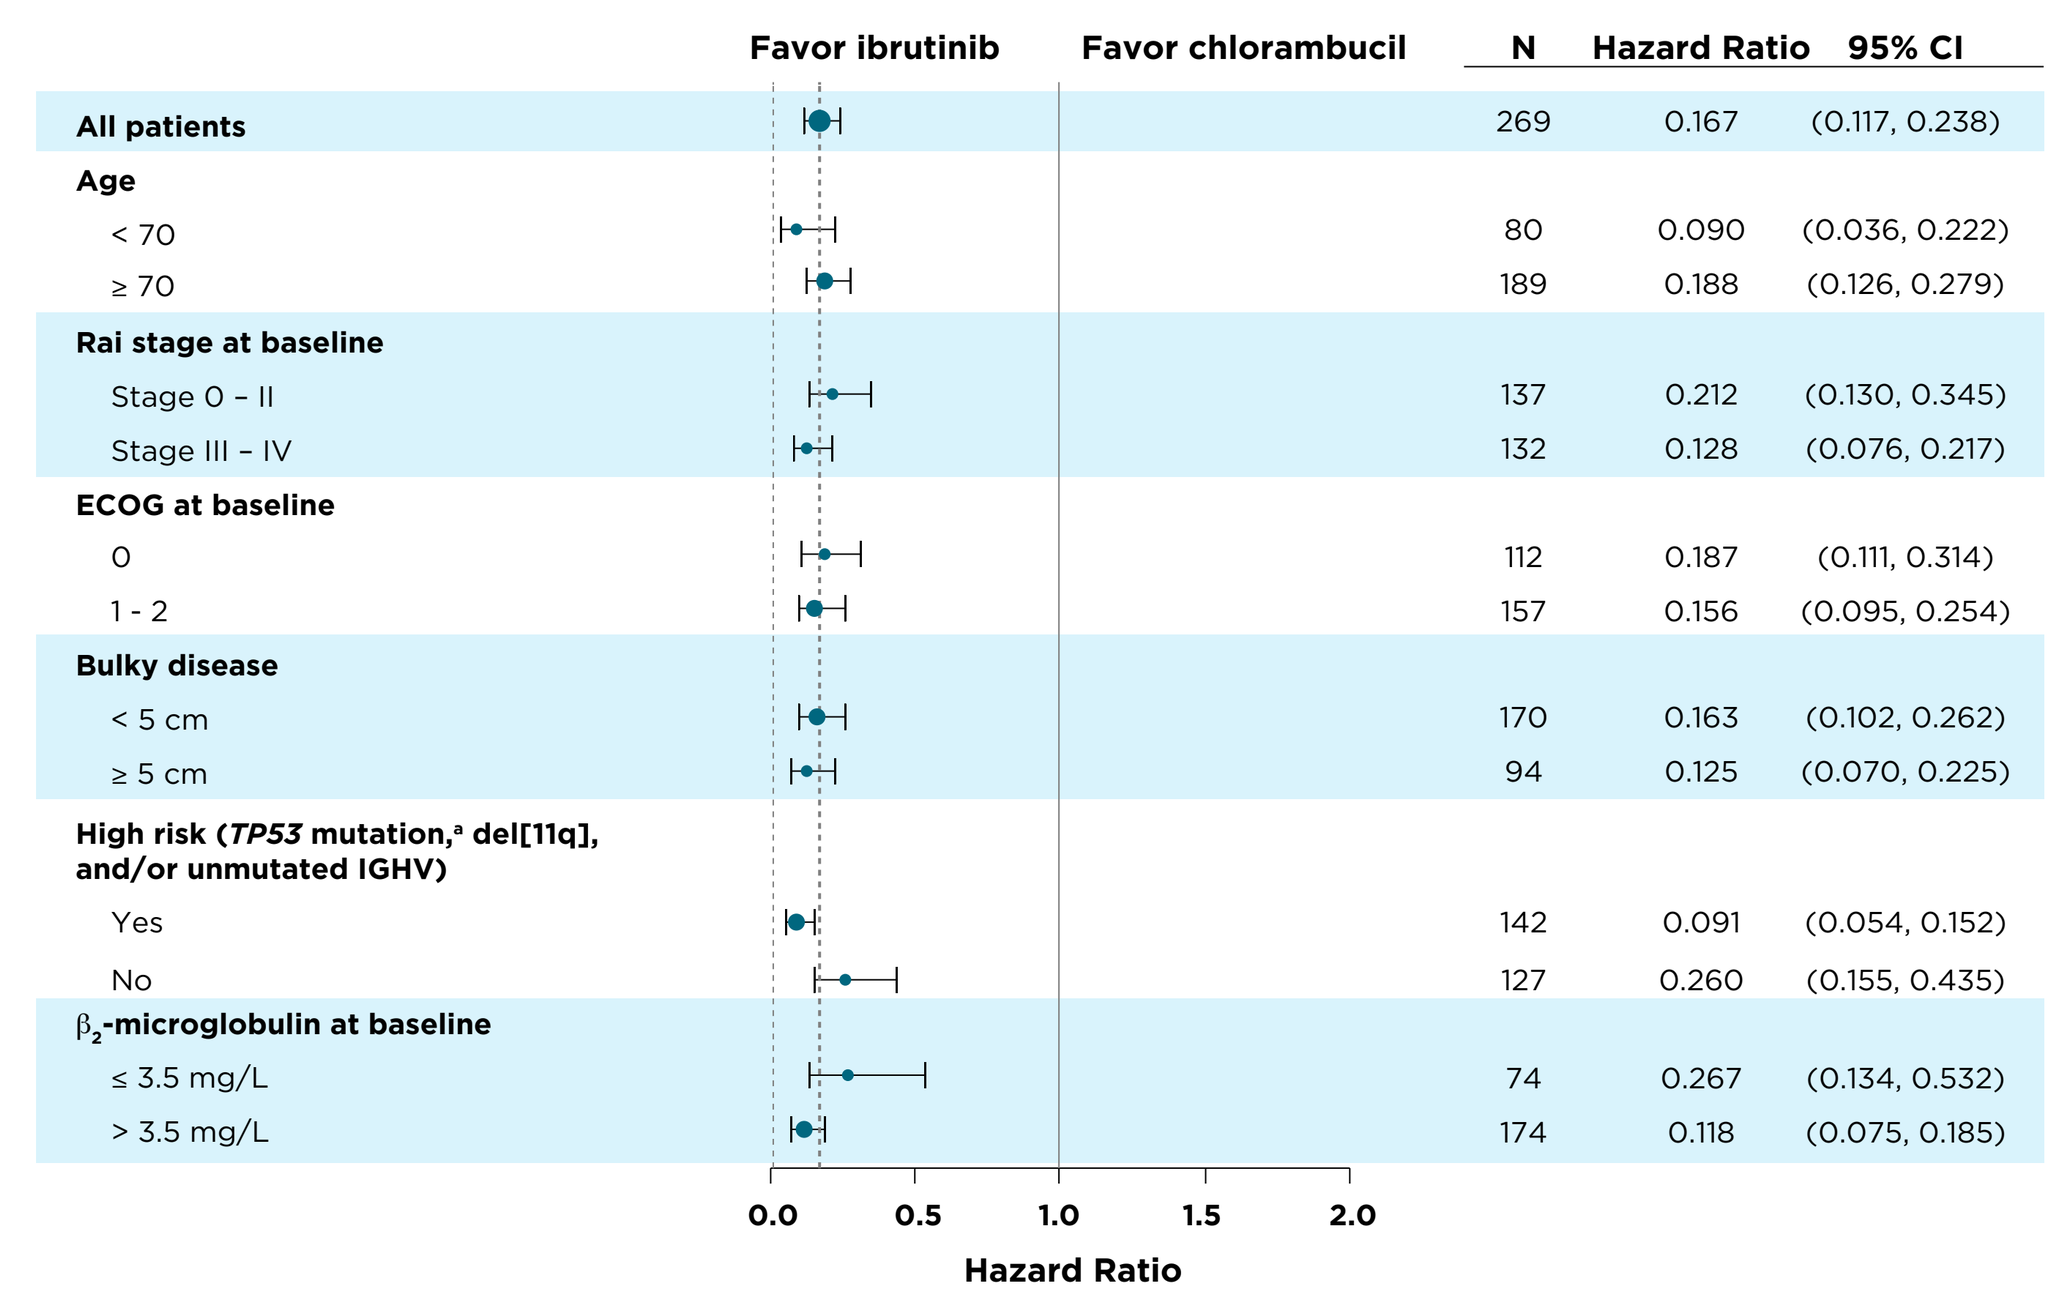
Favor ibrutinib
Favor chlorambucil
N Hazard Ratio 95% CI
All patients 269 0.167 (0.117, 0.238)
Age
< 70 80 0.090 (0.036, 0.222)
≥ 70 189 0.188 (0.126, 0.279)
Rai stage at baseline
Stage 0 – II 137 0.212 (0.130, 0.345)
Stage III – IV 132 0.128 (0.076, 0.217)
ECOG at baseline
0 112 0.187 (0.111, 0.314)
1 - 2 157 0.156 (0.095, 0.254)
Bulky disease
< 5 cm 170 0.163 (0.102, 0.262)
≥ 5 cm 94 0.125 (0.070, 0.225)
High risk (TP53 mutation,[a] del[11q], and/or unmutated IGHV)
Yes 142 0.091 (0.054, 0.152)
No 127 0.260 (0.155, 0.435)
β2-microglobulin at baseline
≤ 3.5 mg/L 74 0.267 (0.134, 0.532)
> 3.5 mg/L 174 0.118 (0.075, 0.185)
Hazard Ratio

[a]TP53 mutation per arm: ibrutinib (n=11) and chlorambucil (n=3).
HR from stratified Cox regression model.

Figure 4. PFS by del(11q) Status

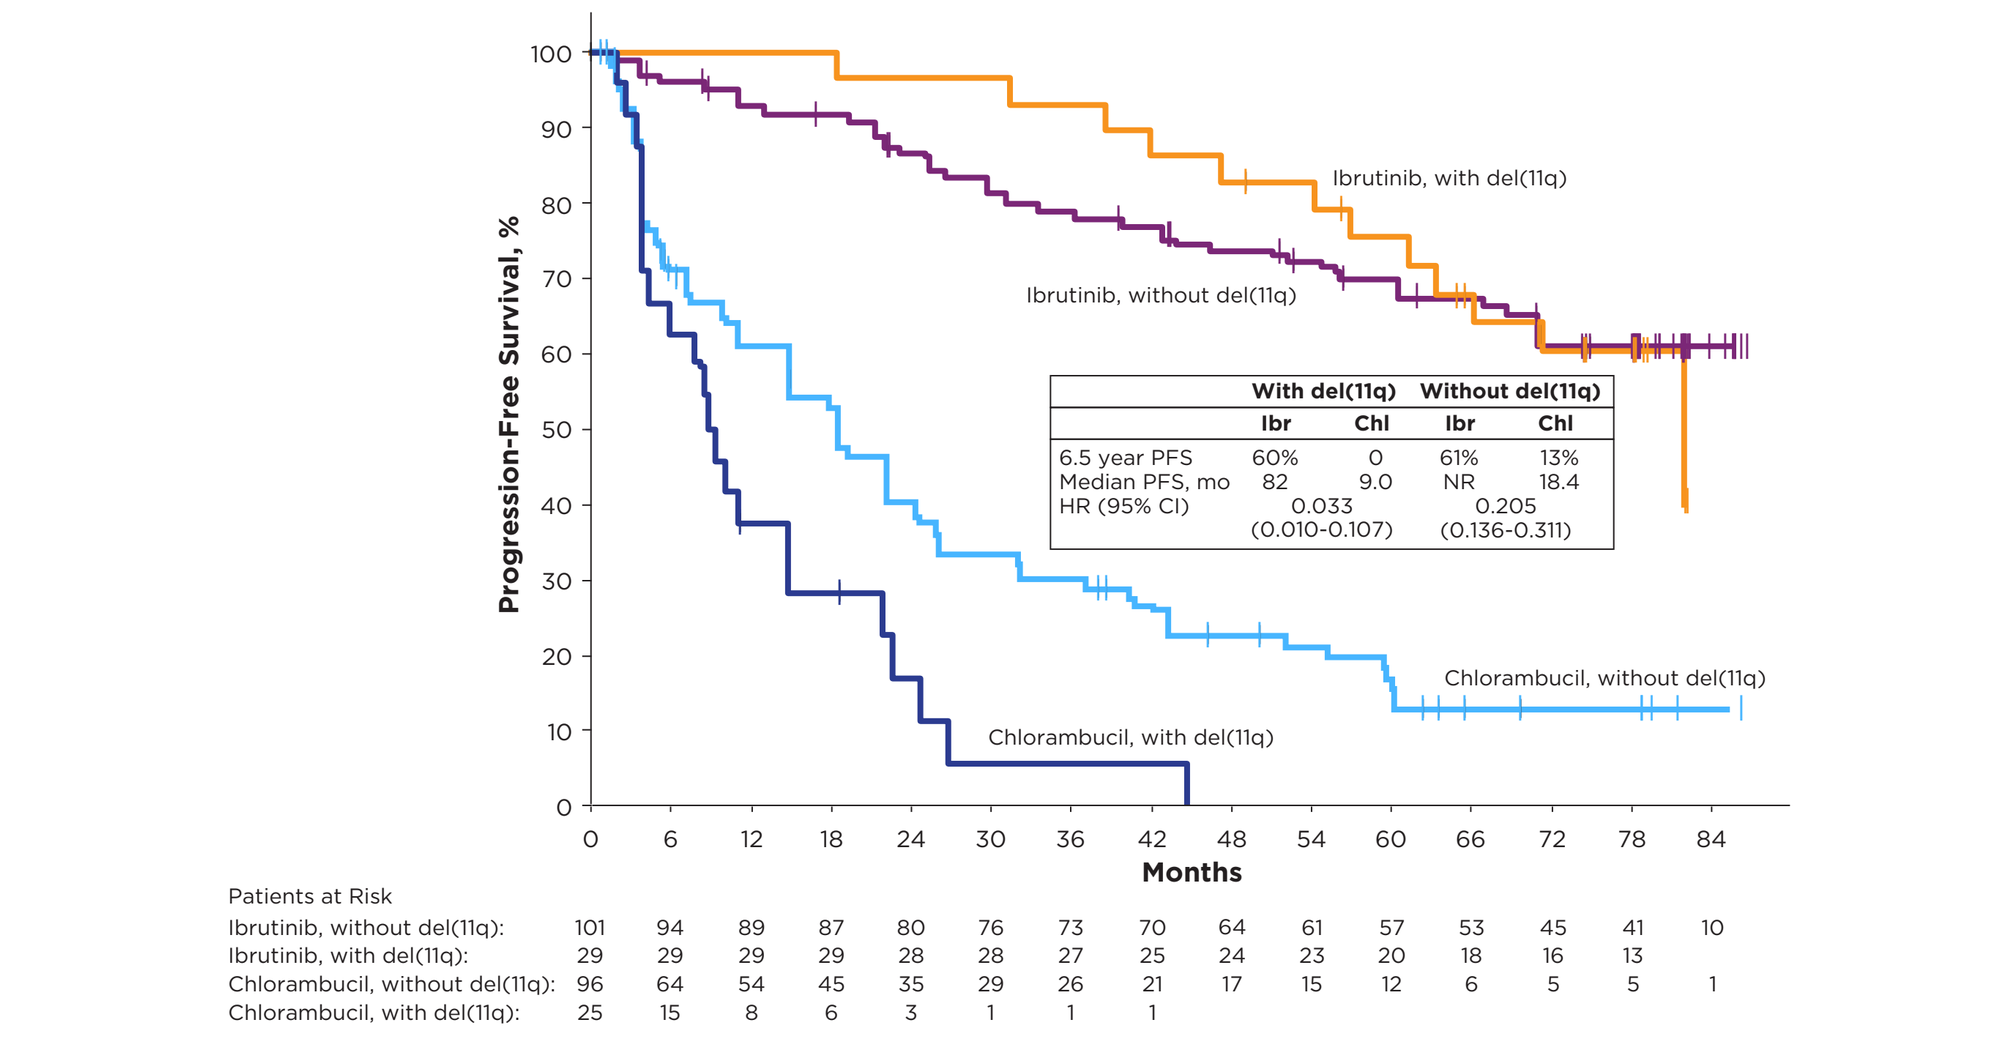
Progression-Free Survival, %
Ibrutinib, with del(11q)
Ibrutinib, without del(11q)
Chlorambucil, without del(11q)
Chlorambucil, with del(11q)
With del(11q) Without del(11q)
Ibr Chl Ibr Chl
6.5 year PFS 60% 0 61% 13%
Median PFS, mo 82 9.0 NR 18.4
HR (95% CI) 0.033 (0.010-0.107) 0.205 (0.136-0.311)
Months
Patients at Risk
Ibrutinib, without del(11q): 101 94 89 87 80 76 73 70 64 61 57 53 45 41 10
Ibrutinib, with del(11q): 29 29 29 29 28 28 27 25 24 23 20 18 16 13
Chlorambucil, without del(11q): 96 64 54 45 35 29 26 21 17 15 12 6 5 5 1
Chlorambucil, with del(11q): 25 15 8 6 3 1 1 1

HR from stratified Cox regression model; artifact at the end of the curve for ibrutinib patients with del(11q) is due to the extremely low patient numbers at these time points.

- Ibrutinib led to a 97% reduction in risk of PD or death in patients with del(11q) and 80% for those without del(11q) vs chlorambucil (**Figure 4**).
- At 6.5 years, PFS rates for ibrutinib vs chlorambucil for patients with del(11q) were 60% vs 0%, and without del(11q) were 61% vs 13%.
- PFS was similar in ibrutinib-treated patients with and without del(11q).

Figure 5. PFS by IGHV Status

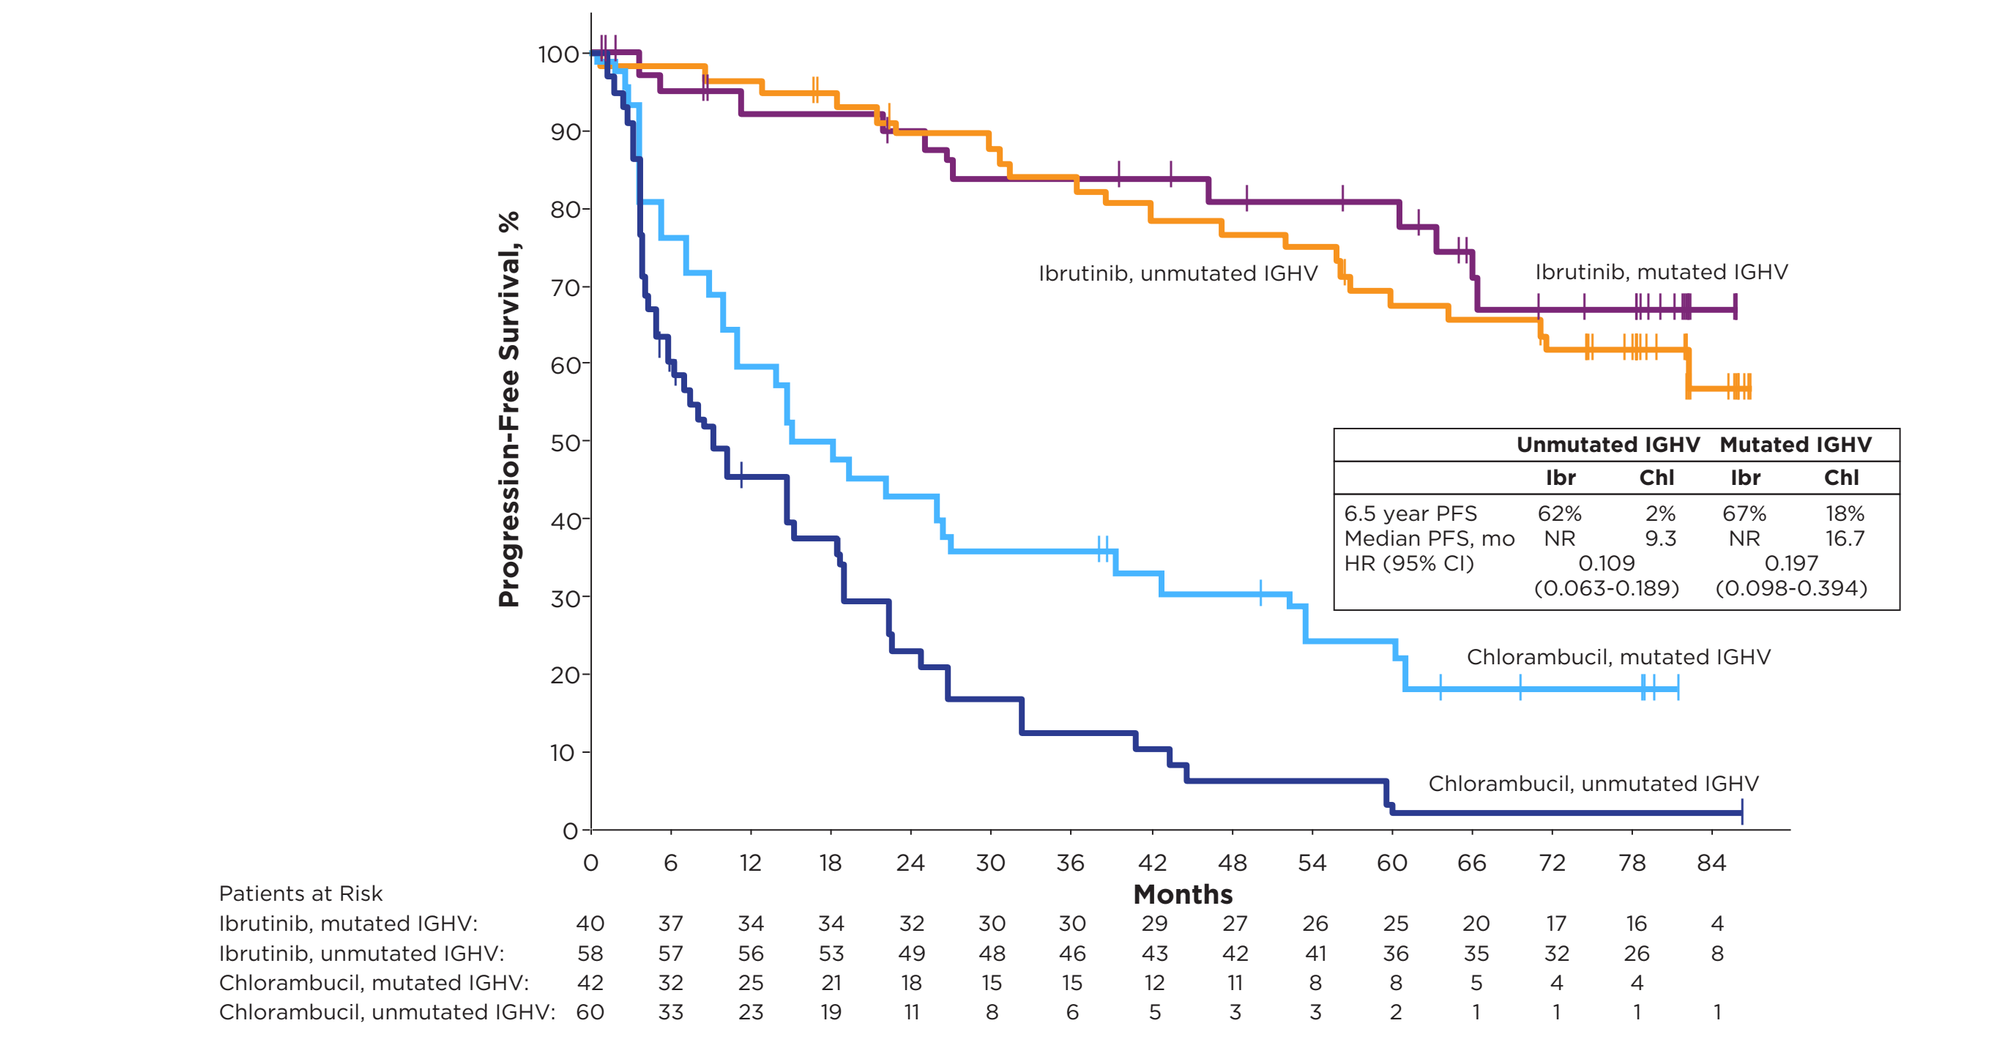
Progression-Free Survival, %
Ibrutinib, unmutated IGHV
Ibrutinib, mutated IGHV
Chlorambucil, mutated IGHV
Chlorambucil, unmutated IGHV
Unmutated IGHV Mutated IGHV
Ibr Chl Ibr Chl
6.5 year PFS 62% 2% 67% 18%
Median PFS, mo NR 9.3 NR 16.7
HR (95% CI) 0.109 (0.063-0.189) 0.197 (0.098-0.394)
Months
Patients at Risk
Ibrutinib, mutated IGHV: 40 37 34 34 32 30 30 29 27 26 25 20 17 16 4
Ibrutinib, unmutated IGHV: 58 57 56 53 49 48 46 43 42 41 36 35 32 26 8
Chlorambucil, mutated IGHV: 42 32 25 21 18 15 15 12 11 8 8 5 4 4
Chlorambucil, unmutated IGHV: 60 33 23 19 11 8 6 5 3 3 2 1 1 1 1

HR from stratified Cox regression model.

- Ibrutinib led to 89% and 80% reductions in the risk of PD or death in patients with unmutated and mutated IGHV, respectively, vs chlorambucil (**Figure 5**).
- At 6.5 years, PFS rates for ibrutinib vs chlorambucil for patients with unmutated IGHV were 62% vs 2%, and with mutated IGHV were 67% vs 18%.
- PFS was similar in ibrutinib-treated patients with unmutated and mutated IGHV.

Figure 6. Investigator-Assessed Overall Response Rate With Ibrutinib

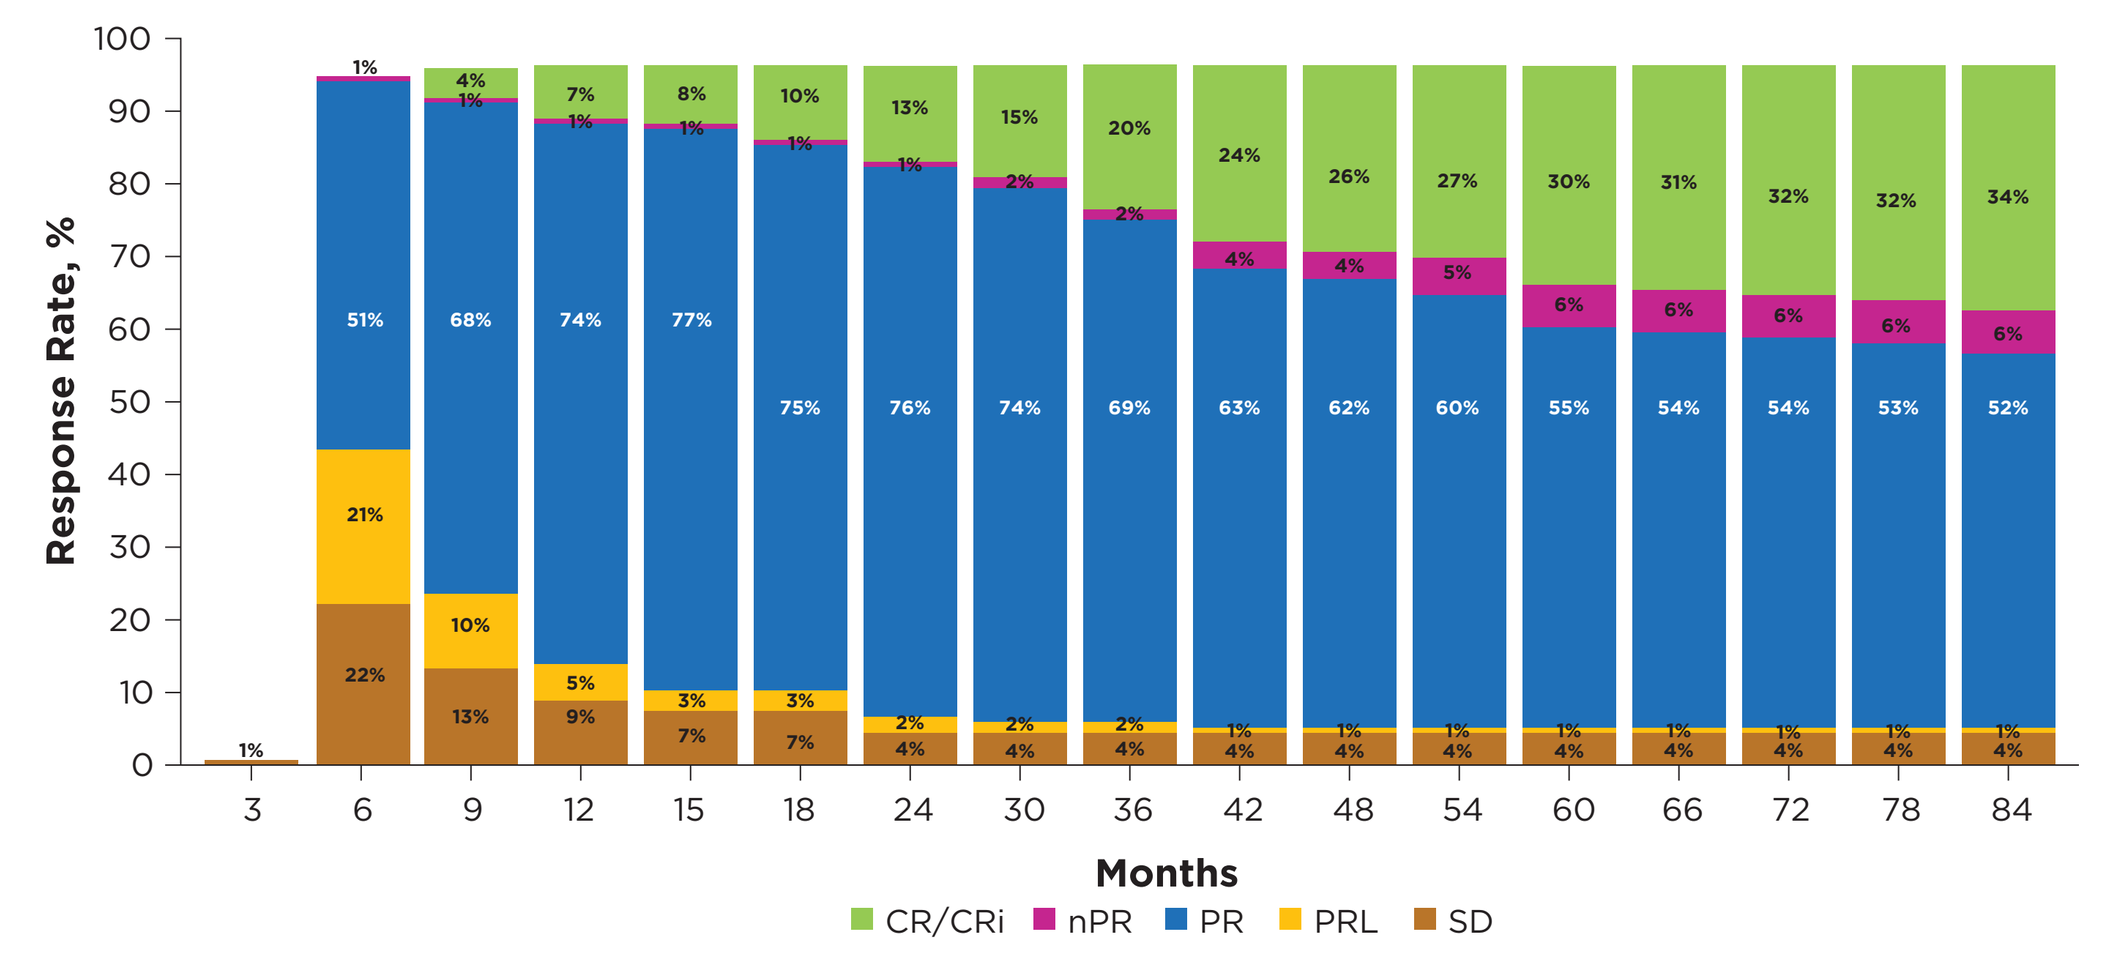
Response Rate, %
Months
CR/CRi nPR PR PRL SD

CR, complete response; CRi, complete response with incomplete bone marrow recovery; nPR, nodular partial response; PR, partial response; PR-L, partial response with lymphocytosis; SD, stable disease.

- At up to 7 years of follow-up, the CR/CRi rate increased over time to 34% (ORR 92%) (**Figure 6**).
 - Median duration of response (range, <0.1 to 83) and complete response (range, <0.1 to 79 months) were not reached.
- For ibrutinib-treated patients, ORR and CR rates were similar, irrespective of del(11q) or IGHV mutation status.

Figure 7. Ibrutinib OS

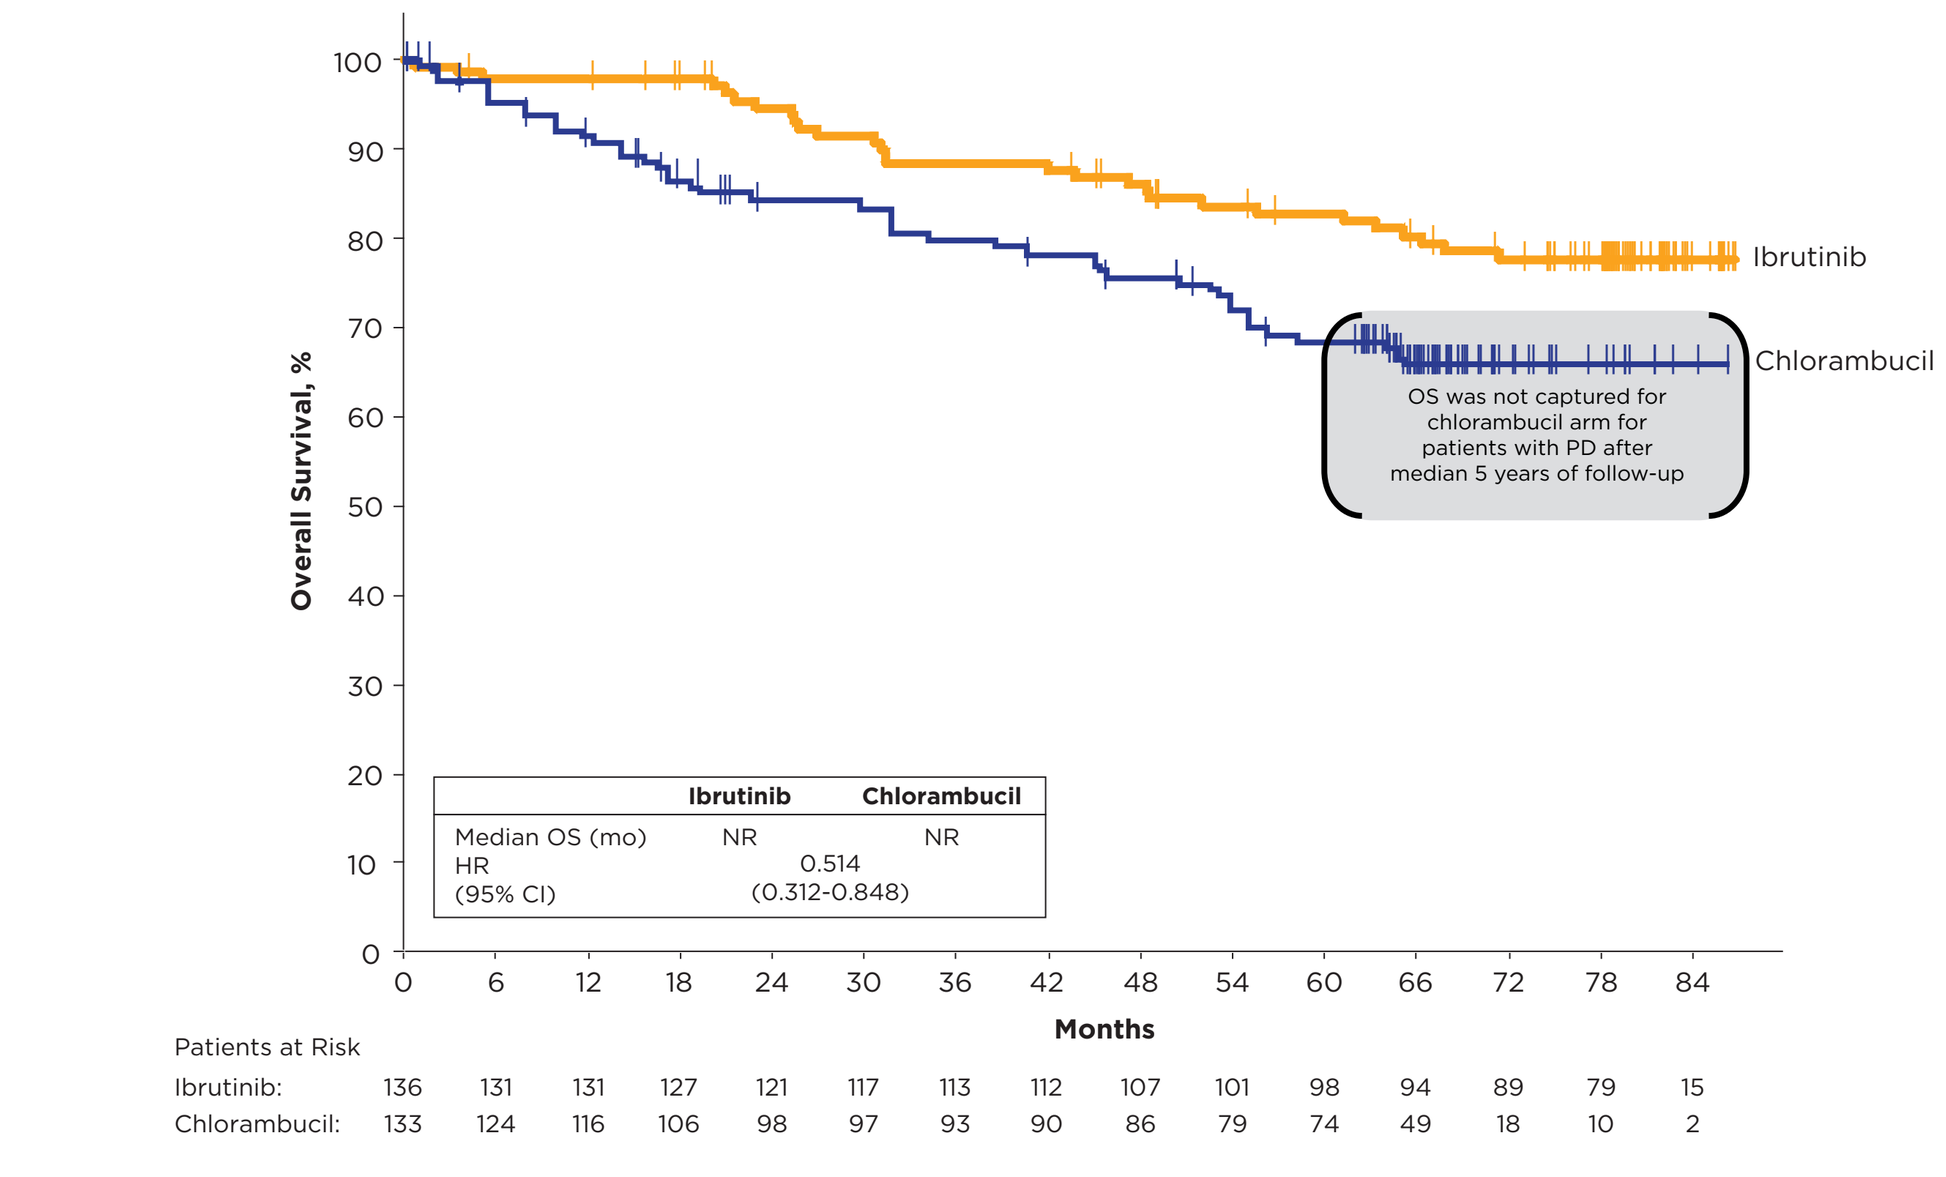
Overall Survival, %
Ibrutinib
Chlorambucil
OS was not captured for chlorambucil arm for patients with PD after median 5 years of follow-up
Ibrutinib Chlorambucil
Median OS (mo) NR NR
HR (95% CI) 0.514 (0.312-0.848)
Months
Patients at Risk
Ibrutinib: 136 131 131 127 121 117 113 112 107 101 98 94 89 79 15
Chlorambucil: 133 124 116 106 98 97 93 90 86 79 74 49 18 10 2

HR from stratified Cox regression model.

- Patients treated with ibrutinib had improved OS and a 49% reduction in death (**Figure 7**).
- 78% of ibrutinib-treated patients were estimated to be alive at 6.5 years.
 - OS estimates at 5 years for ibrutinib vs chlorambucil were 83% vs 68%.
- Patients with high-risk genomic features (*TP53* mutation, del[11q], or unmutated IGHV) had similar improved OS with ibrutinib treatment as observed for all patients (HR: 0.489 [95% CI, 0.249–0.960]).

Safety

Table 1. Ibrutinib Treatment Disposition

	First-Line Ibrutinib (n=136[a])
Median duration of ibrutinib treatment, years[b] (range)	6.2 (0.06–7.2)
Continuing ibrutinib on study, n (%)	64 (47)
Discontinued ibrutinib, n (%)	
AE	31 (23)
PD	16 (12)
Death	11 (8)
Withdrawal by patient	9 (7)
Investigator decision	4 (3)

[a]One patient received no doses of ibrutinib; [b]74.0 months (0.7–86.8).
AE, adverse event.

- More than half of patients continued to receive single-agent ibrutinib for at least 6 years.
- 47% of patients continue first-line ibrutinib treatment on study with this follow-up (**Table 1**).

Table 2. Concomitant Medications of Clinical Interest

	Ibrutinib n=135	Chlorambucil n=132
Antithrombotics, n (%)	96 (71)	71 (54)
Antiplatelets	80 (59)	67 (51)
Anticoagulants	48 (36)	13 (10)
Antihypertensives,[a] n (%)	97 (72)	80 (61)
Acid related disorders, n (%)	88 (65)	55 (42)
H_2-receptor antagonists	23 (17)	10 (8)
Proton pump inhibitors	75 (56)	47 (36)
Other[b]	25 (19)	5 (4)
Neutrophil Growth Factors, n (%)	13 (10)	16 (12)

[a]Including antihypertensives, agents acting on the renin-angiotensin system, beta blocking agents, calcium channel blockers; [b]Excluding proton pump inhibitors or H_2-blocker.

- **Table 2** shows concomitant medications of clinical interest by treatment arm.
- Over half of patients on study received concomitant medication for acid-related disorders (54%), including 46% who received proton pump inhibitors.

Figure 8. Prevalence[a] of Most Frequent AEs Over Time in Ibrutinib-Treated Patients

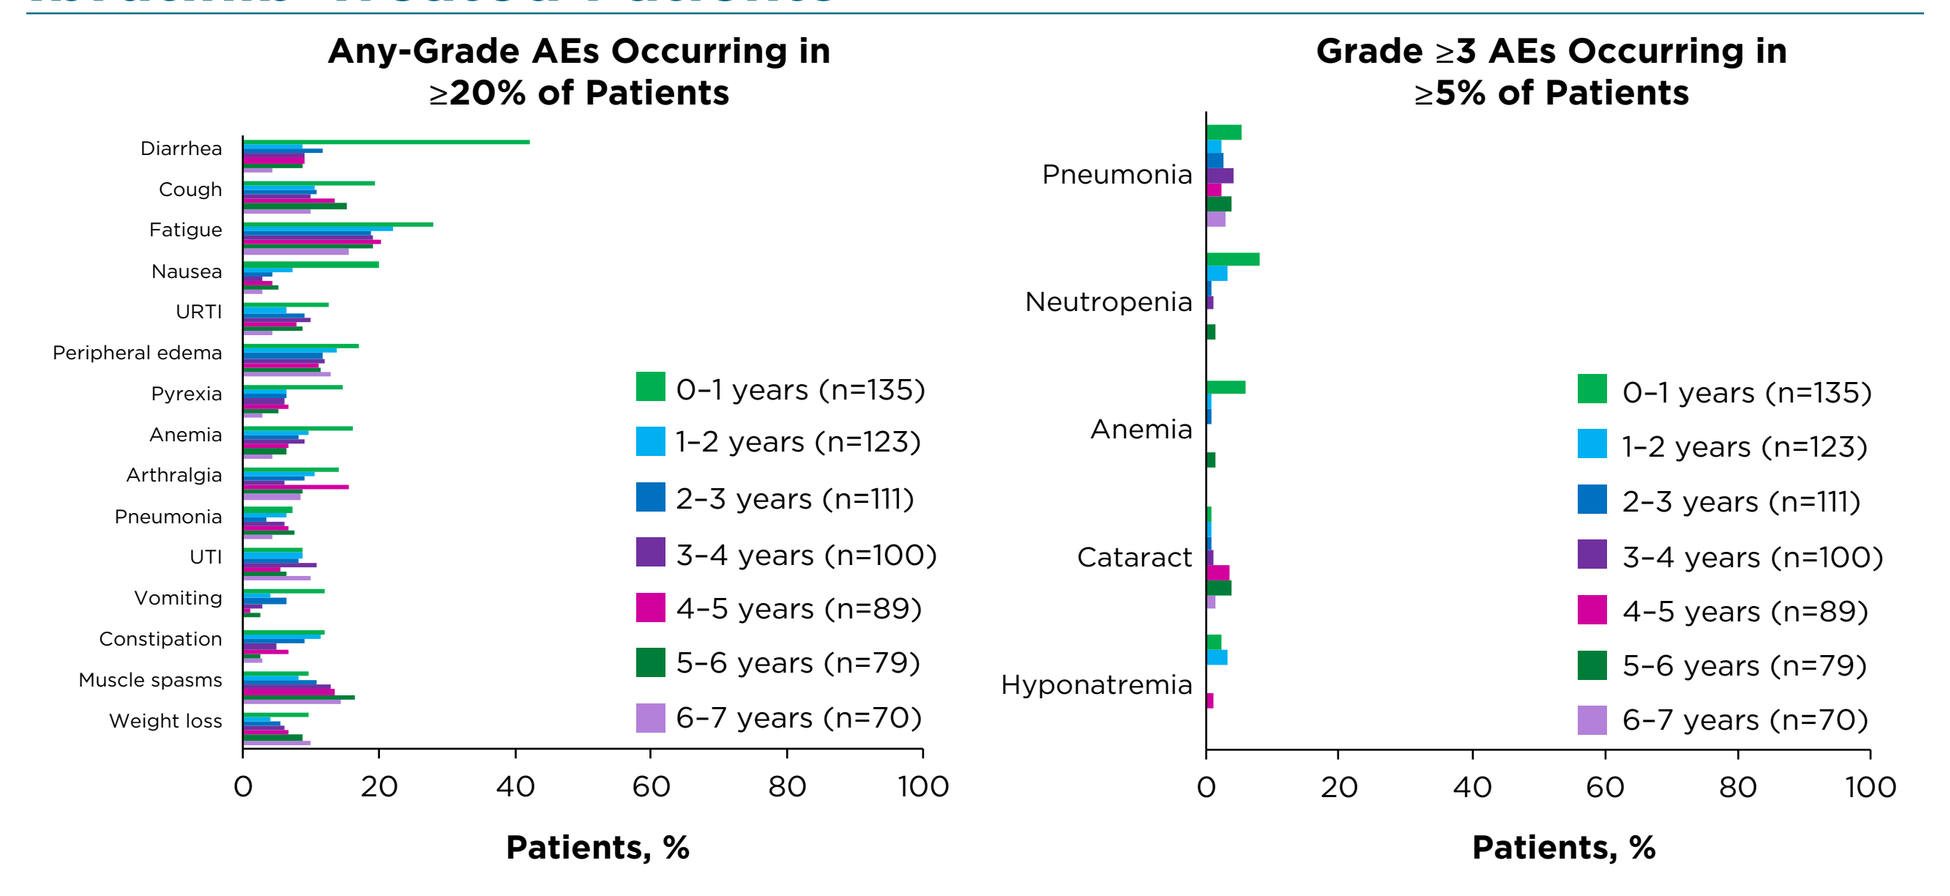
Any-Grade AEs Occurring in ≥20% of Patients
Diarrhea
Cough
Fatigue
Nausea
URTI
Peripheral edema
Pyrexia
Anemia
Arthralgia
Pneumonia
UTI
Vomiting
Constipation
Muscle spasms
Weight loss
0–1 years (n=135)
1–2 years (n=123)
2–3 years (n=111)
3–4 years (n=100)
4–5 years (n=89)
5–6 years (n=79)
6–7 years (n=70)
Patients, %
Grade ≥3 AEs Occurring in ≥5% of Patients
Pneumonia
Neutropenia
Anemia
Cataract
Hyponatremia
Patients, %

[a]Prevalence was determined by the proportion of patients with a given AE (existing event or new onset of an event) during each yearly interval. Multiple onsets of the same AE term within a specific yearly interval were counted once, and the same AE term continuing across several yearly intervals was counted in each of the intervals.
Atrial fibrillation and hypertension are shown in Figure 9, AEs of clinical interest.

- The most frequent any-grade and grade ≥3 AEs are shown (**Figure 8**).

Figure 9. AEs of Clinical Interest Over Time in Patients Treated With Ibrutinib

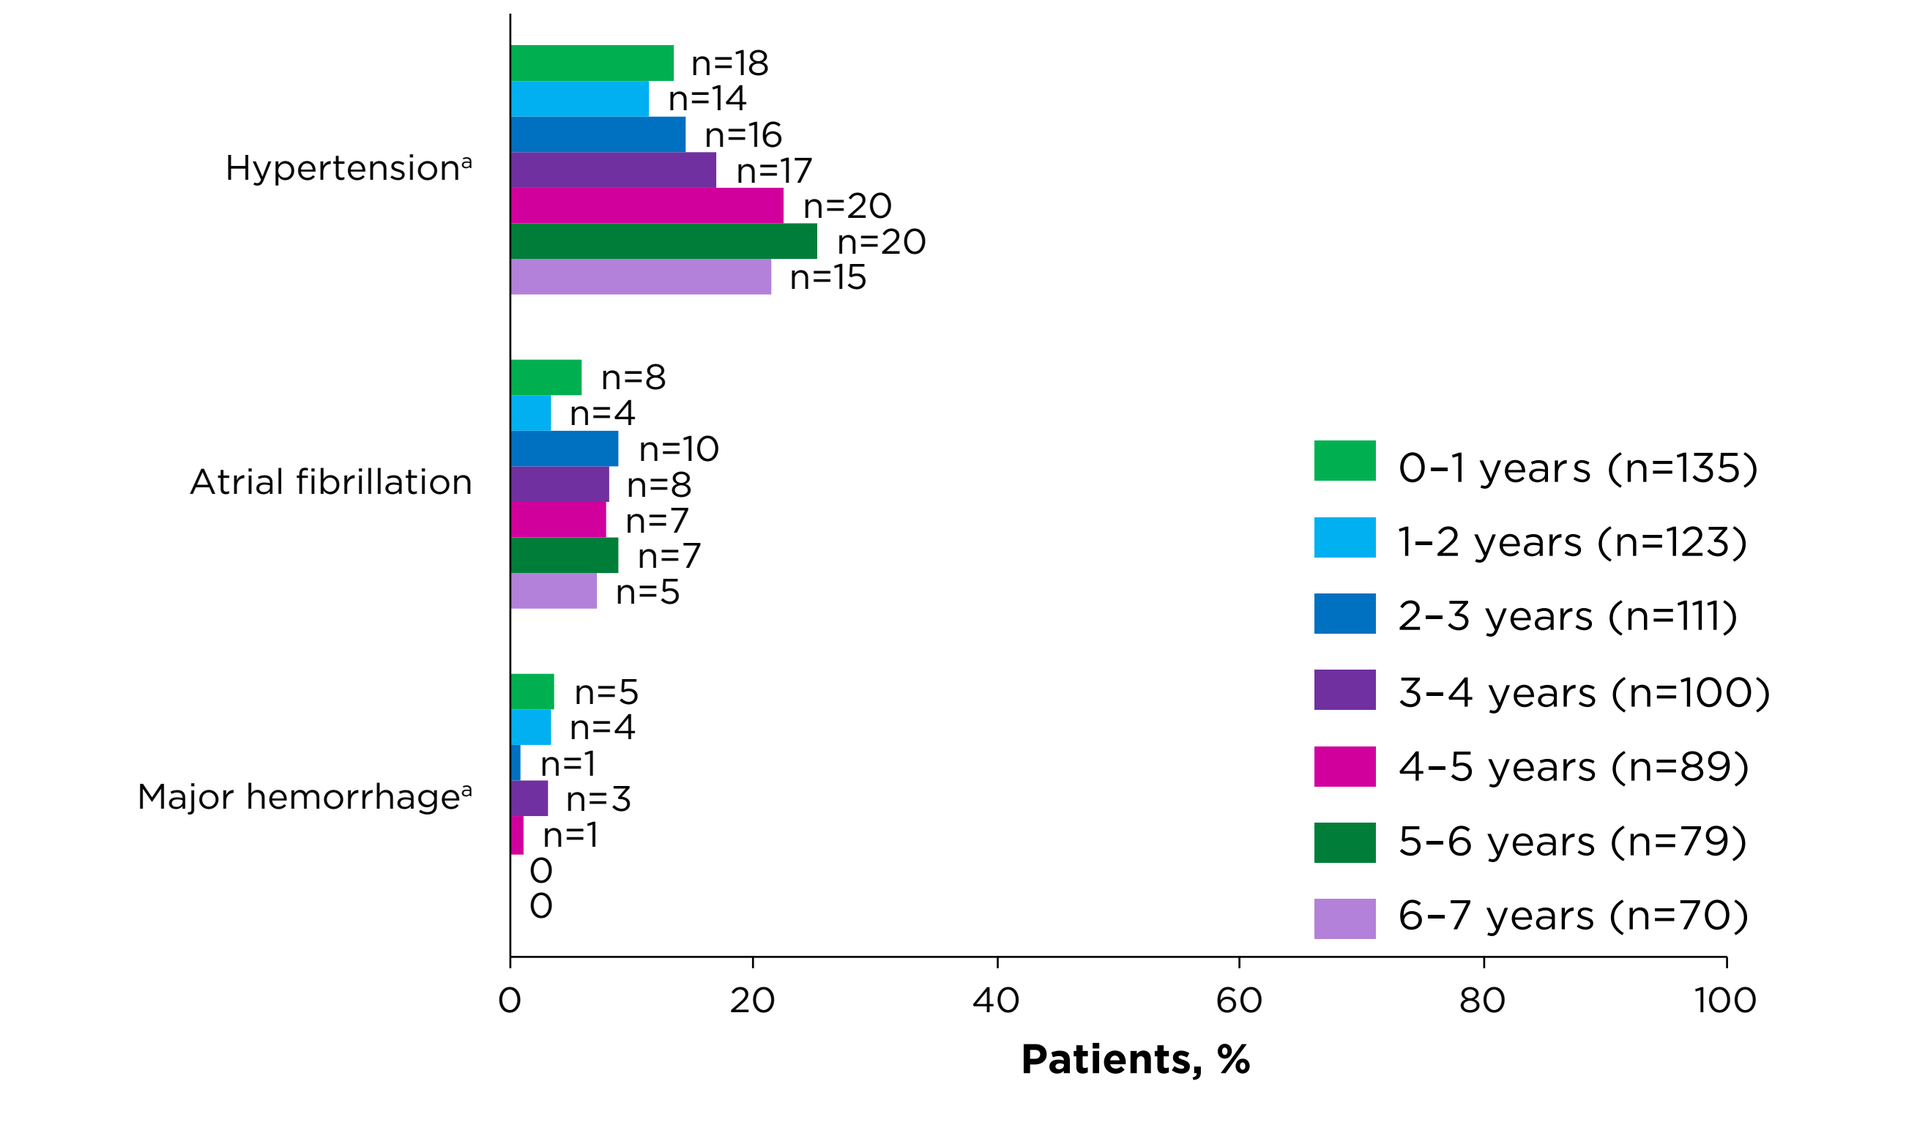
Hypertension[a]
n=18
n=14
n=16
n=17
n=20
n=20
n=15
Atrial fibrillation
n=8
n=4
n=10
n=8
n=7
n=7
n=5
Major hemorrhage[a]
n=5
n=4
n=1
n=3
n=1
0
0
0–1 years (n=135)
1–2 years (n=123)
2–3 years (n=111)
3–4 years (n=100)
4–5 years (n=89)
5–6 years (n=79)
6–7 years (n=70)
Patients, %

[a]Combined terms.

- Prevalence rates of any-grade AEs of clinical interest are shown (**Figure 9**).

Figure 10. AEs Leading to Ibrutinib Dose Reductions or Discontinuations

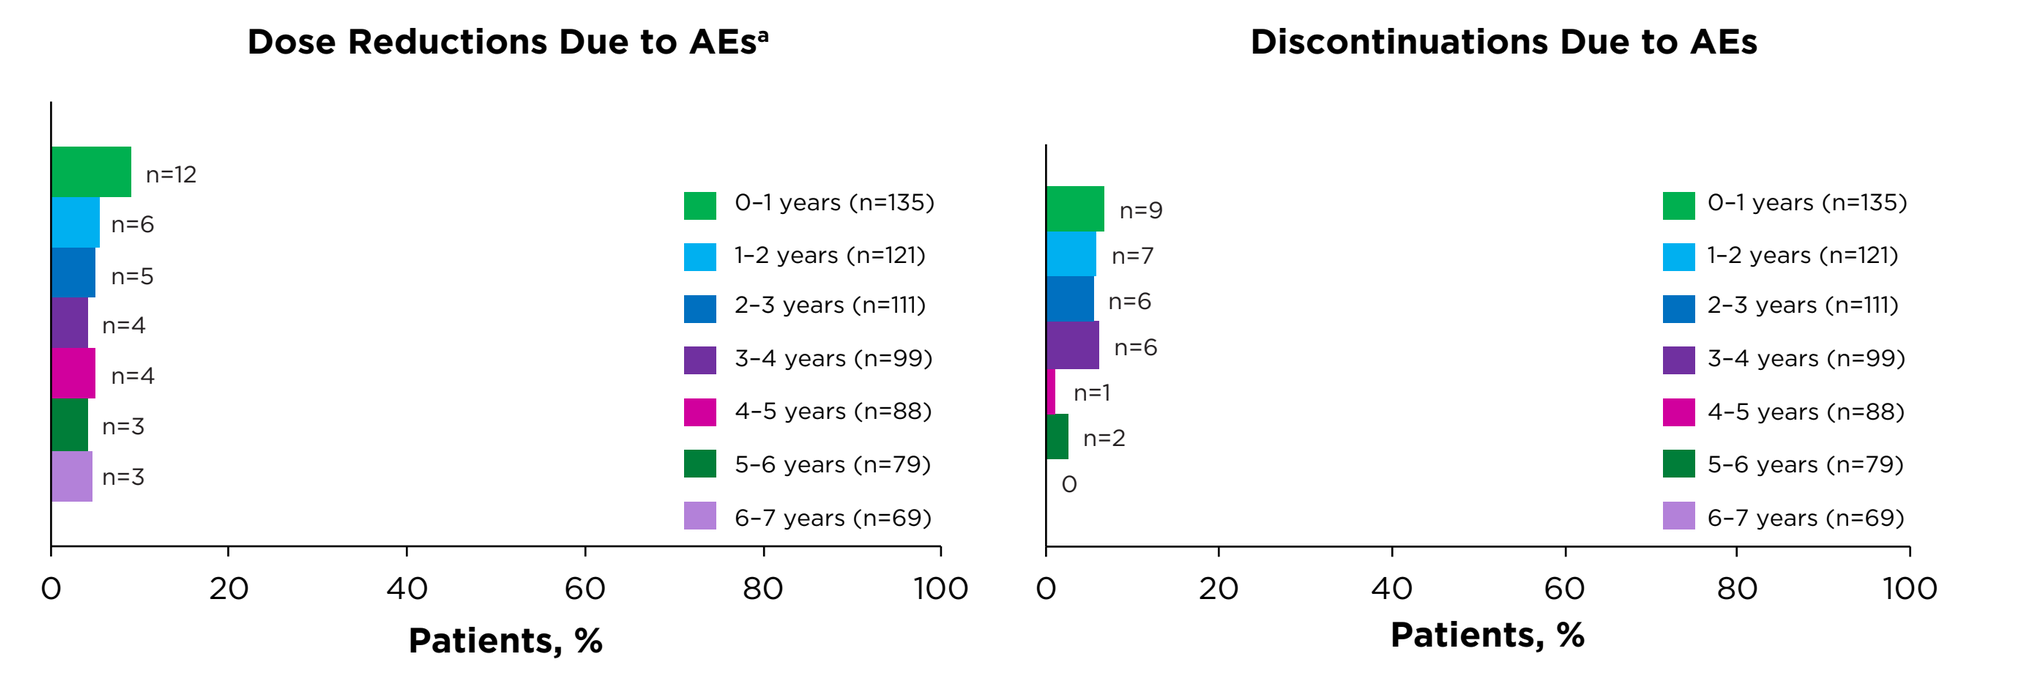
Dose Reductions Due to AEs[a]
n=12
n=6
n=5
n=4
n=4
n=3
n=3
0–1 years (n=135)
1–2 years (n=121)
2–3 years (n=111)
3–4 years (n=99)
4–5 years (n=88)
5–6 years (n=79)
6–7 years (n=69)
Patients, %
Discontinuations Due to AEs
n=9
n=7
n=6
n=6
n=1
n=2
0
Patients, %

[a]The same patient may appear in more than one year.

- Ibrutinib dose reductions and discontinuations due to AEs are shown over time (**Figure 10**).
- 66/79 patients (84%) had an AE that had a complete resolution following a dose hold of at least 7 days.
- 31 patients (23%) experienced AEs leading to dose reductions.
 - AEs occurring in >1 patient were thrombocytopenia (n=3), and anemia, arthralgia, diarrhea, fatigue, and palpitations (n=2, each).
 - 22/31 (71%) had subsequent AE improvement or resolution.
- At current follow-up (up to 7 years), 31 patients (23%) experienced AEs as the primary cause of ibrutinib discontinuation.
 - AEs occurring in >1 patient were atrial fibrillation (n=5), pneumonia (n=3), and palpitations (n=2).

Table 3. Outcomes Following Ibrutinib Discontinuation

	Discontinued Due to AEs n=31	Discontinued Due to PD n=16	Discontinued Due to Any Cause Other Than Death n=60
Median (range) time on study after discontinuation, months	35.6 (0.2-81.5)	18.7 (<0.1- 53.9)	21.5 (<0.1-81.5)
Median (95% CI) OS after discontinuation, months	NR (15.7–NE)	NR (1–NE)	NR (26.3–NE)
Alive,[a] n (%)	21 (68)	11 (69)	43 (72)

[a]Includes patients who exited study with no known death.

- First subsequent therapies reported after ibrutinib discontinuation (n=19) included chemoimmunotherapy (n=9; FCR, BR, GC, or vincristine+R), chemotherapy (n=2), novel agents (n=7; including 4 patients on venetoclax), and radiation (n=1).
 - An additional three patients received venetoclax or venetoclax+R as their second subsequent or later line of therapy after ibrutinib.
- Best overall response to the first subsequent drug therapy was reported for 10/18 patients to date: 7/10 responded, 1/10 had stable disease, and 2/10 had PD.

CONCLUSIONS

- In the longest follow-up to date (up to 7 years) from a phase 3 study of first-line BTK-directed therapy, single-agent ibrutinib conferred sustained PFS and OS benefit vs chlorambucil.
 - With ibrutinib treatment, median investigator-assessed PFS and OS were not reached and 6.5-year estimates were 61% and 78%, respectively.
 - Only 16 (12%) patients progressed while receiving ibrutinib.
 - PFS benefit for ibrutinib vs chlorambucil was similar in patients with high-risk genomic features (*TP53* mutation, del[11q], and/or unmutated IGHV) and in lower-risk patients.
 - With up to 7 years of follow-up, ibrutinib benefit was similar for patients with mutated or unmutated IGHV.
 - Responses continued to deepen over time, with further increase in CR/CRi rates with ibrutinib treatment.
- Ibrutinib remains well tolerated with no new safety signals observed with long-term follow-up.
 - Active dose management (dose holds and reductions) to address AEs enabled most patients who required dose management to continue benefiting from ibrutinib treatment.
 - With extended follow-up, rates of discontinuations due to AEs remain low.
- Nearly half of patients remain on ibrutinib at up to 7 years of follow-up.

REFERENCES

1. IMBRUVICA [prescribing information]. Sunnyvale, CA: Pharmacyclics LLC; 2020.
2. Byrd, *N Eng J Med*. 2014;371(3):213-23.
3. Burger, *N Eng J Med*. 2015;373(25):2425-37.
4. Moreno, *Lancet Oncol*. 2018;20(1):43-56.
5. Woyach, *N Eng J Med*. 2018;379(26):2517-28.
6. Shanafelt, *N Eng J Med*. 2019;381(5):432-43.
7. Fraser, *Leukemia*. 2019;33(4):969-80.
8. Winqvist, *Haematologica*. 2016;101(12):1573-80.
9. UK CLL Forum, *Haematologica*. 2016;101(12):1563-72.
10. Mato, *Ann Oncol*. 2017;28(5):1050-6.
11. Mato, *Haematologica*. 2018;103(5):874-9.
12. Mato, *Am J Hematol*. 2018;93(11)1394-401.
13. Winqvist, *Haematologica*. 2019;104(5):e208-e210.
14. Mauro, ASH 2018. Abs 3147.
15. Janssens, EHA 2019. Abs PF384.
16. Aarup, ASH 2019. Abs 1767.
17. Salles, *Ann Hematol*. 2019;98(12):2749-60.
18. Van der Straten, *Br J Haematol*. 2020;188(6):e109-e112.
19. Pula, EHA 2020. Abs PB1904.
20. Abrisqueta, EHA 2020. Abs EP710.
21. Burger, *Leukemia*. 2020;34(3):787-98.

DISCLOSURES

JAB: honoraria from and consultancy/advisory role with Janssen; research funding from AstraZeneca, BeiGene, and Pharmacyclics LLC, an AbbVie Company; speakers' bureau for and travel expenses from Gilead, Janssen, Novartis, Pharmacyclics LLC, an AbbVie Company, and TG Therapeutics; **PMB:** consultancy/advisory role with AbbVie, AstraZeneca, Bristol Myers Squibb, Celgene, Genentech, Gilead, Janssen, MEI Pharma, Merck, Morphosys, MS Pharma, Pharmacyclics LLC, an AbbVie Company, Seattle Genetics, and TG Therapeutics; research funding from AstraZeneca and TG Therapeutics; **CO:** honoraria from AbbVie, AstraZeneca, Gilead, Incyte, Janssen, Merck, Roche, and Teva; **TR:** honoraria from AstraZeneca and Janssen; consultancy/advisory role with and research funding from Acerta, AstraZeneca and Janssen; **AT:** honoraria from and speakers' bureau for AbbVie, AstraZeneca, BeiGene, and Janssen; **OB:** consultancy/advisory role with AbbVie, AstraZeneca, and Janssen; research funding from Janssen; **PH:** consultancy/advisory role with AbbVie, AstraZeneca, and Janssen; research funding from AbbVie, AstraZeneca, Gilead, Janssen, Novartis/GSK, Pharmacyclics LLC, an AbbVie Company, and Roche; speakers' bureau for AbbVie, AstraZeneca, Janssen, and Roche; travel expenses from AbbVie and Janssen; **SEC:** honoraria from AbbVie, Janssen, and Pharmacyclics LLC, an AbbVie Company; consultancy/advisory role with AbbVie, Adaptive, AstraZeneca, BeiGene, Celgene, Genentech, Janssen, Novartis, and Pharmacyclics LLC, an AbbVie Company; research funding from AbbVie, AstraZeneca, Janssen, and Pharmacyclics LLC, an AbbVie Company; expert testimony for Genentech and Janssen; **SD, SG, JL & FO:** no relevant disclosures; **HM:** honoraria and travel expenses from Janssen; consultancy/advisory role with AbbVie and Janssen; **CM:** consultancy/advisory role with AbbVie, AstraZeneca, BeiGene, and Janssen; research funding from AbbVie and Janssen; speakers' bureau for Janssen; **SD:** employment with Pharmacyclics LLC, an AbbVie Company, and Horizon Therapeutics; stock ownership in AbbVie, Bristol Myers Squibb, Exelixis, Gilead, GlaxoSmithKline, Horizon Therapeutics, Myovant Sciences, and Revance Therapeutics; **AS & JPD:** employment with Pharmacyclics LLC, an AbbVie Company; stock ownership in AbbVie; **TJK:** employment with Moores Cancer Center; stock ownership in Oncternal; honoraria from AbbVie, Celgene, DAVA Oncology, Genentech, Gilead, Janssen, Pharmacyclics LLC, an AbbVie Company, and Roche; consultancy/advisory role with AbbVie, Celgene, DAVA Oncology, Genentech-Roche, Gilead, Janssen, and Pharmacyclics LLC, an AbbVie Company; research funding from Celgene, Crim, MD Anderson Cancer Center, Oncternal Therapeutics, Pharmacyclics LLC, an AbbVie Company, and Velos; speakers' bureau for AbbVie, DAVA Oncology, Genentech, Gilead, Janssen, Pharmacyclics LLC, an AbbVie Company, and Verastem; patents, royalties, or other intellectual property with the University of California and Oncternal Therapeutics, Inc., in the development and licensing of cirmtuzumab; travel expenses from AbbVie, Bionest Partners, Celgene, DAVA Oncology, Genentech, Gilead, Indy Hematology Review, Janssen, OncLive, Pharmacyclics LLC, an AbbVie Company, Roche, TG Therapeutics, and Verastem; **PG:** honoraria from and consultancy/advisory role with AbbVie, Acerta/AstraZeneca, Adaptive, ArQule/MSD, BeiGene, Celgene/Juno Therapeutics/Bristol Myers Squibb, Gilead, Janssen, Lilly/Loxo, and Roche; research funding from AbbVie, AstraZeneca, Gilead, Janssen, Novartis, and Sunesis.

ACKNOWLEDGMENTS

We thank the patients who participated in the study and their supportive families, as well as the investigators and clinical research staff from the study centers. We acknowledge David Simpson, MBChB, FRACP, FRCPA, for his contribution to the study. This study was sponsored by Pharmacyclics LLC, an AbbVie Company. Editorial support was provided by Emily Chastain, PhD, an employee of Pharmacyclics LLC, an AbbVie Company.

To submit a medical question, please visit www.pharmacyclicsmedinfo.com

Poster presented at the Society of Hematologic Oncology (SOHO) Annual Meeting, September 8-11, 2021, Houston, TX