AML-162

Efficacy and Safety of Venetoclax in Combination With Gilteritinib for Relapsed/Refractory FLT3-Mutated Acute Myeloid Leukemia: Updated Analyses of a Phase 1b Study

Jessica K Altman[1], Naval Daver[2], Joseph Maly[3], Mark Levis[4], Ellen Ritchie[5], Mark Litzow[6], James McCloskey[7], Catherine C Smith[8], Gary Schiller[9], Terrence Bradley[10,11], Ramon V Tiu[12], Kiran Naqvi[13], Satya Siddani[14], Jing Wang[14], Paul Lee[14], Alexander E Perl[15]

[1]Robert H. Lurie Comprehensive Cancer Center, Northwestern University, Chicago, Illinois, United States; [2]Department of Leukemia, University of Texas MD Anderson Cancer Center, Houston, Texas, United States; [3]Norton Cancer Institute, Louisville, Kentucky, United States; [4]Sidney Kimmel Comprehensive Cancer Center, Johns Hopkins University, Baltimore, Maryland, United States; [5]Division of Hematology and Oncology, Weill Cornell Medical College, New York, New York, United States; [6]Division of Hematology, Mayo Clinic, Rochester, Minnesota, United States; [7]John Theurer Cancer Center at Hackensack Meridian Health, Hackensack, New Jersey, United States; [8]Helen Diller Family Comprehensive Cancer Center, University of California San Francisco, San Francisco, California, United States; [9]David Geffen School of Medicine at UCLA, Los Angeles, California, United States; [10]University of Miami, Department of Medicine, Miami, Florida, United States; [11]Sylvester Comprehensive Cancer Center, University of Miami, Miami, Florida, United States; [12]Astellas Pharma US, Northbrook, Illinois, United States; [13]Genentech, South San Francisco, California, United States; [14]AbbVie Inc., North Chicago, Illinois, United States; [15]Abramson Cancer Center, University of Pennsylvania, Philadelphia, Pennsylvania, United States

Presented at Society of Hematologic Oncology 2021 (SOHO) • September 8–11, 2021 • Houston, Texas

BACKGROUND

- The FMS-like tyrosine kinase 3 (FLT3) tyrosine kinase inhibitor (TKI) gilteritinib (Gilt) improved survival in relapsed/refractory (R/R) FLT3-mutated (*FLT3*$^{mut+}$) acute myeloid leukemia (AML)[1]
- However, relapse was common and long-term survival remained short in the phase 3 ADMIRAL trial (median overall survival [OS]: 9.3 mo, 2-year OS 20%)[2,3]
- 88% of patients treated on ADMIRAL had not received a prior FLT3 TKI[2]
- Combination of FLT3 TKIs with B-cell lymphoma 2 (BCL-2) inhibitors demonstrated preclinical synthetic lethality[4]
- Venetoclax (Ven) is a selective BCL-2 inhibitor; approved in combination with hypomethylating agents or low dose cytarabine for the treatment of newly diagnosed AML not suitable for induction therapy[5]

OBJECTIVE

- This multicenter, open-label, phase 1b clinical trial aims to assess the efficacy and safety of Ven in combination with Gilt for R/R *FLT3*$^{mut+}$ AML (NCT03625505)

METHODS

Figure 1. Study Design

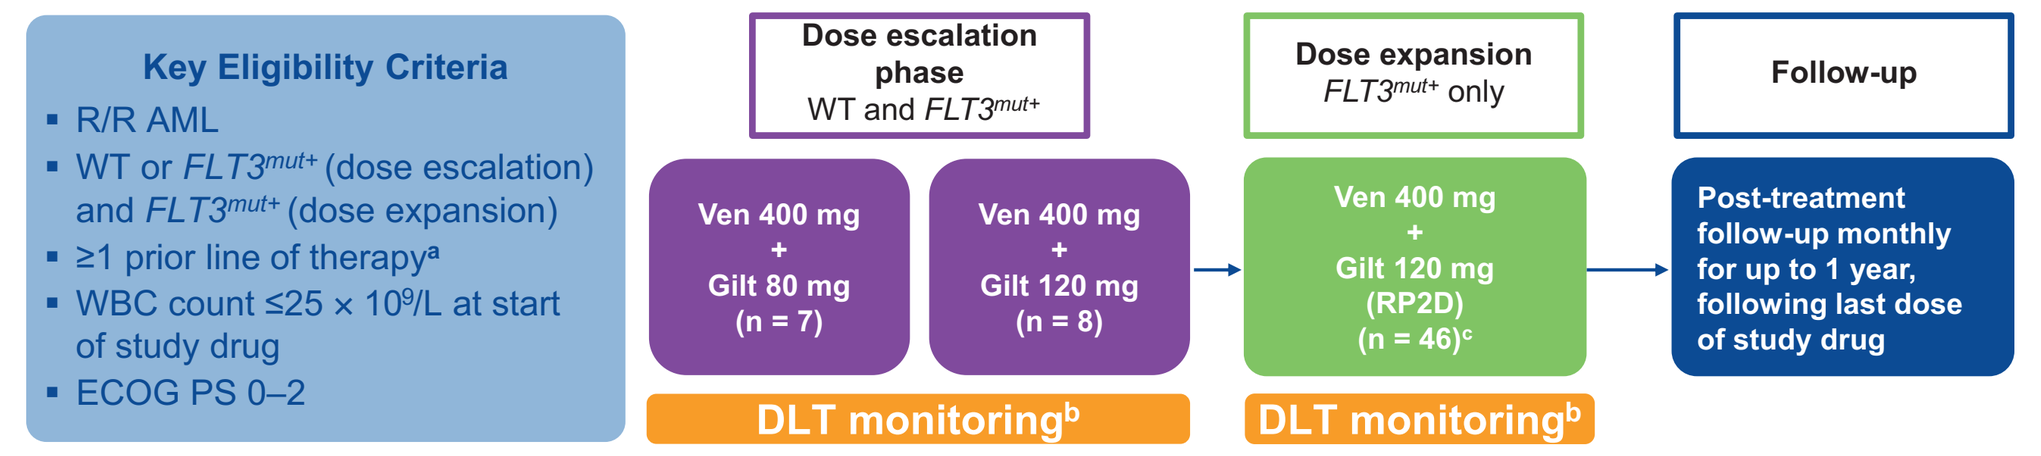
Key Eligibility Criteria
R/R AML
WT or FLT3mut+ (dose escalation) and FLT3mut+ (dose expansion)
≥1 prior line of therapya
WBC count ≤25 × 10⁹/L at start of study drug
ECOG PS 0–2
Dose escalation phase
WT and FLT3mut+
Ven 400 mg + Gilt 80 mg (n = 7)
Ven 400 mg + Gilt 120 mg (n = 8)
DLT monitoringb
Dose expansion
FLT3mut+ only
Ven 400 mg + Gilt 120 mg (RP2D) (n = 46)c
DLT monitoringb
Follow-up
Post-treatment follow-up monthly for up to 1 year, following last dose of study drug

Primary Endpoints	Secondary Endpoints	Exploratory Endpoints
• mCRc[d]	• CR + CRh • DoR of mCRc	• OS in *FLT3*$^{mut+}$ subgroups • Allelic burden of FLT3 mutations

[a]Prior exposure to Ven was permitted in a protocol amendment (unless received as part of a randomized controlled trial); FLT3 inhibitors including Gilt were allowed during dose escalation; FLT3 inhibitors except Gilt were permitted during dose expansion; prior stem cell transplant was permitted. [b]DLT evaluation period defined as first 28 days of Cycle 1. [c]Of the N = 56 *FLT3*$^{mut+}$ patients presented 46 were from the expansion phase. [d]mCRc, modified composite complete remission, defined as: complete response (CR) + CR with incomplete platelet recovery (CRp) + modified complete response with incomplete neutrophil count recovery (CRi*) + morphologic leukemia-free state (MLFS), by modified IWG criteria for AML, which were adapted from IWG criteria to match the "CRc" criteria used in the phase 3 ADMIRAL study of Gilt, to allow comparison with the CRc used in Gilt development. The IWG criteria of CRi are different from the criteria adopted by investigators and modified to have IWG response CRi* that is mutually exclusive with IWG response CRp. Hematology Criteria for CRi*: ANC ≤1 × 10⁹/L and platelet >100 × 10⁹/L.
DoR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; OS, overall survival; RP2D, recommended phase 2 dose; WT, wild type.

RESULTS

- 62.5 % of patients received ≥1 prior FLT3 TKI

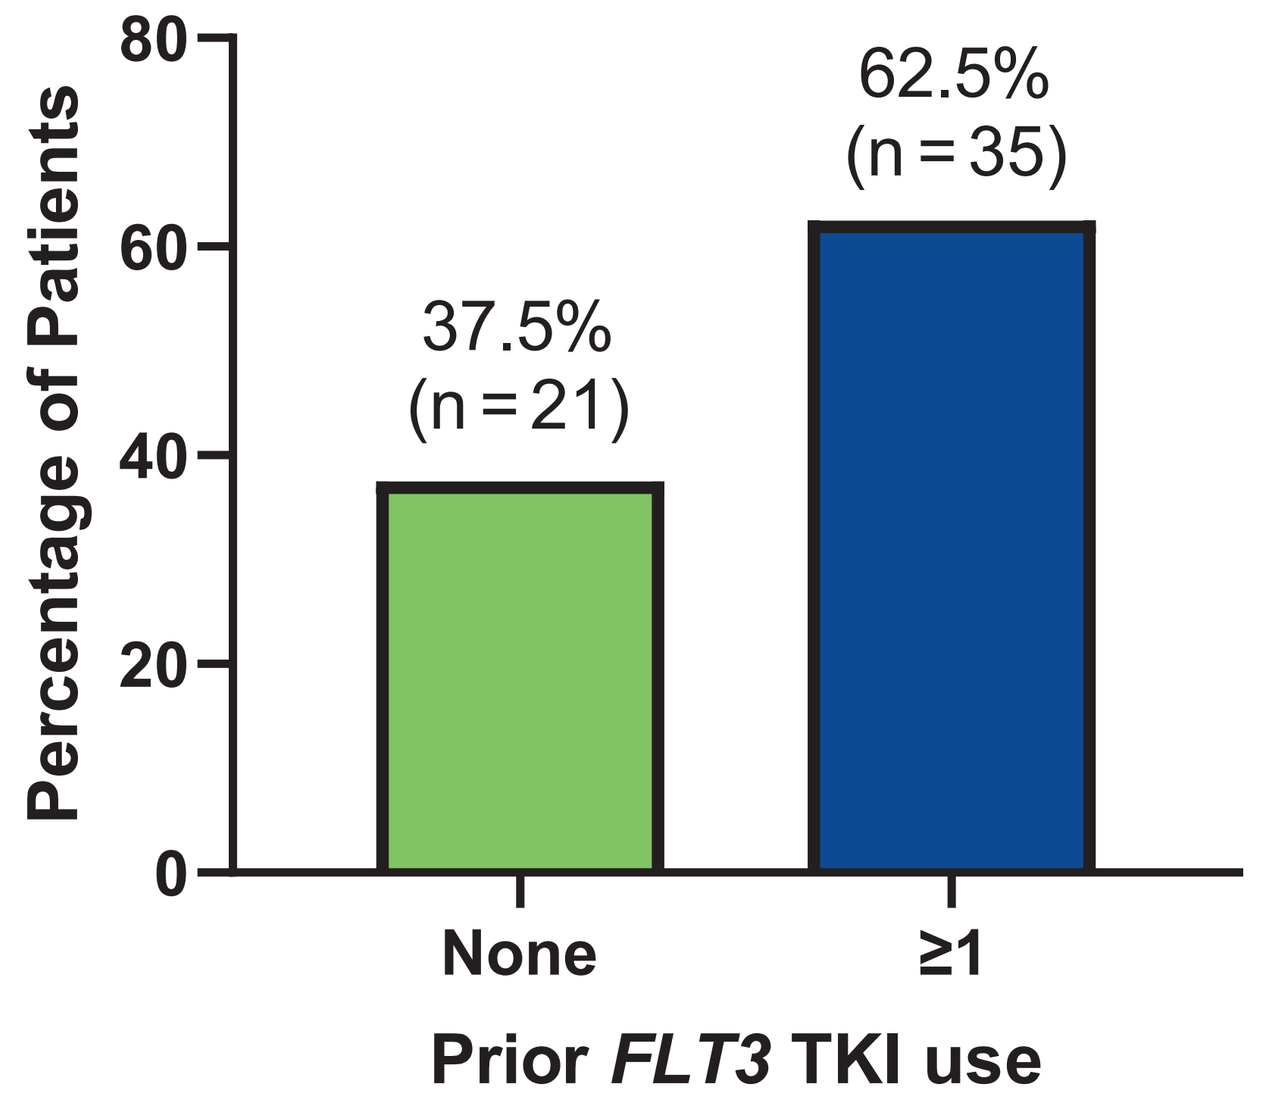
Percentage of Patients
80
60
40
20
0
37.5% (n = 21)
62.5% (n = 35)
None
≥1
Prior FLT3 TKI use

RESULTS (CONTINUED)

Demographic characteristics		All patients at any dose N = 56
Female		29 (51.8)
Age, median (range), years		63 (21, 85)
	Age, ≥60 years, n (%)	32 (57.1)
FLT3 mutation	ITD only	44 (78.6)
	ITD ± TKD	47 (83.9)
	TKD only	9 (16.0)
Cytogenetic risk[a]	Favorable	2 (3.7)
	Intermediate	31 (57.4)
	Poor	17 (31.5)
	No mitoses	4 (7.4)
	Missing	2
AML type	De novo	44 (78.6)
	Secondary	12 (21.4)
Prior lines of therapy	Median (min-max)	2 (1, 5)
	1	13 (23.2)
	2	22 (39.3)
	≥3	21 (37.5)
Prior venetoclax		10 (17.9)
Prior HSCT		18 (32.1)

Data cut off: January 31, 2021; Data are n (%) unless otherwise stated.
[a]Percentages calculated on non-missing values (n = 54).
HSCT, Hematopoietic stem cell transplantation; ITD, internal tandem duplications; TKD, tyrosine kinase domain.

SAFETY PROFILE

- 30-day mortality rate was 0%
- 60-day mortality rate was 10.9% (n = 6)
- 1 adverse event (AE) of clinical TLS
- Dosing modifications due to AE were similar for Ven and Gilt
- Dose interruptions were the most common dosing modification due to AE

n(%)	All patients treated at any dose N = 56
Any AE	56 (100)
Any AE possibly related to Ven	47 (83.9)
Any AE possibly related to Gilt	48 (85.7)
Any AE Grade ≥3	53 (94.6)
Grade 3+ AE cytopenias	44 (78.6)
Any SAE	41 (73.2)
All deaths	28 (50.0)
Fatal AE[a]	10 (17.9)
Clinical TLS	1 (1.8)

n (%)	Ven N = 56	Gilt N = 56
Any AE leading to discontinuation[b]	7 (12.5)	6 (10.7)
Any AE leading to dose reduction[c]	4 (7.1)	4 (7.1)
Any AE leading to dose interruption	30 (53.6)	28 (50.0)

[a]Preferred terms used for AEs leading to death included: Colitis, disease progression, multiple organ dysfunction syndrome, bronchopulmonary aspergillosis, pneumonia, sepsis, systemic mycosis, respiratory failure, subdural hematoma, and malignant neoplasm progression. [b]Febrile neutropenia, ventricular tachycardia, colitis, fall, subdural haematoma, alanine aminotransferase increased, blood bilirubin increased, neutrophil count decreased (all n = 1). [c]Blood bilirubin increased (n = 2); febrile neutropenia, colitis, fall, blood lactate dehydrogenase increased (all n = 1).
AE, adverse event; Gilt, gilteritinib; SAE, serious adverse event; Ven, venetoclax.

SUMMARY OF BEST RESPONSES

- The mCRc rate was 76.4% compared to the CRc rate of 54% (using the same response parameters) in the single agent Gilt ADMIRAL phase 3 study[2]

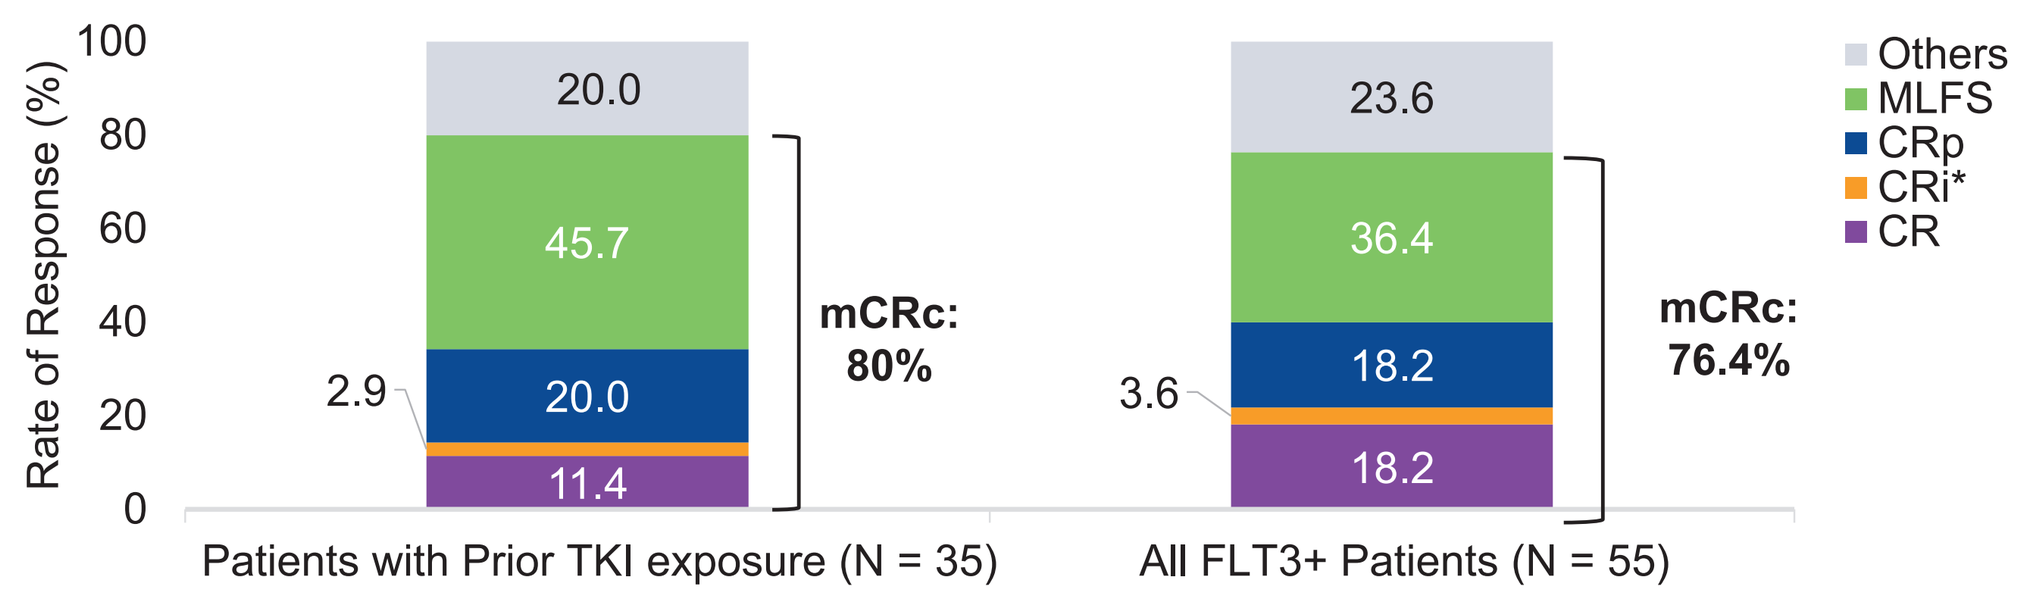
Rate of Response (%)
100
80
60
40
20
0
20.0
45.7
20.0
11.4
2.9
mCRc: 80%
23.6
36.4
18.2
18.2
3.6
mCRc: 76.4%
Others
MLFS
CRp
CRi*
CR
Patients with Prior TKI exposure (N = 35)
All FLT3+ Patients (N = 55)

	FLT3+ patients with prior TKI (N = 35)	All FLT3+ patients (N = 55a)
mCRc, n (%)	28 (80.0)	42 (76.4)
CR+CRp+CRi*	12 (34.3)	22 (40.0)
MLFS	16 (45.7)	20 (36.4)
Time to mCRc (months), median (range)	0.9 (0.7, 4.2)	0.9 (0.7, 4.6)

[a]One FLT3+ patient did not have a post-baseline disease assessment and was therefore not included in these analyses.

OVERALL SURVIVAL, BEST RESPONSE, AND POST-TREATMENT ALLOGENEIC TRANSPLANT

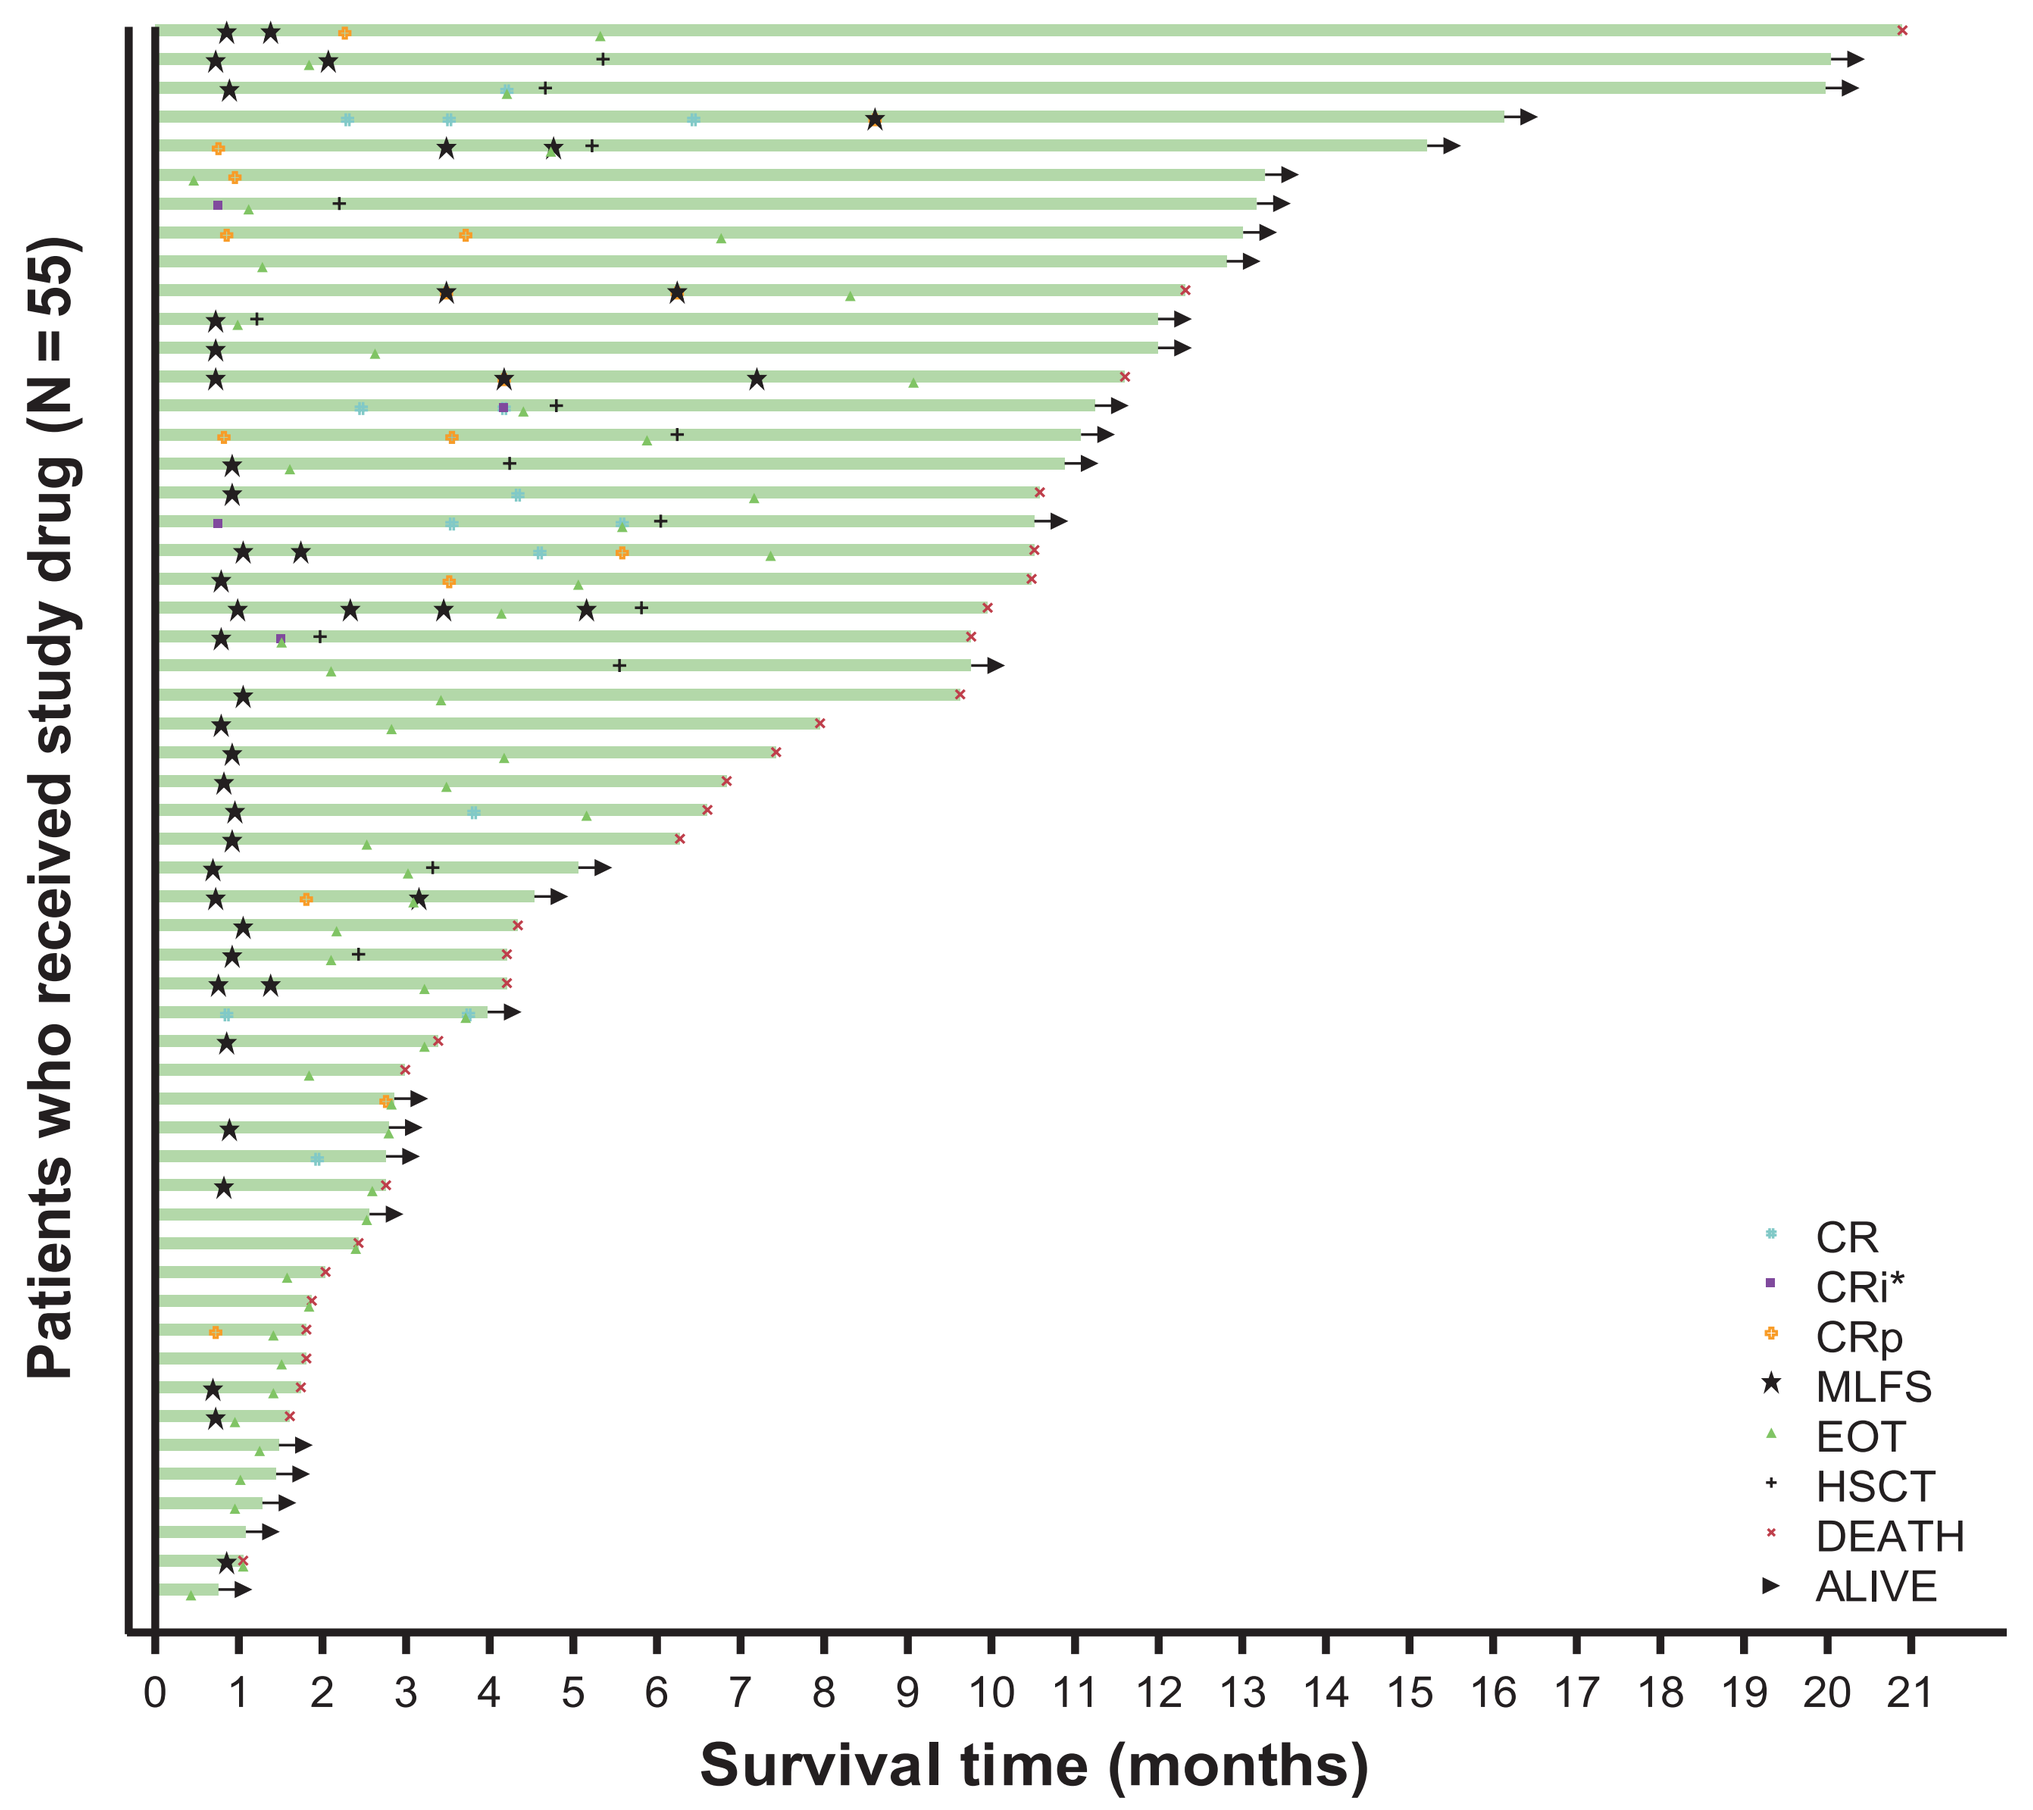
Patients who received study drug (N = 55)
Survival time (months)
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21
CR
CRi*
CRp
MLFS
EOT
HSCT
DEATH
ALIVE

CRp and CRi* are derived from laboratory data.
EOT, patient discontinued all study treatments; HSCT, first post-treatment allogeneic transplant.

DURATION OF mCRc

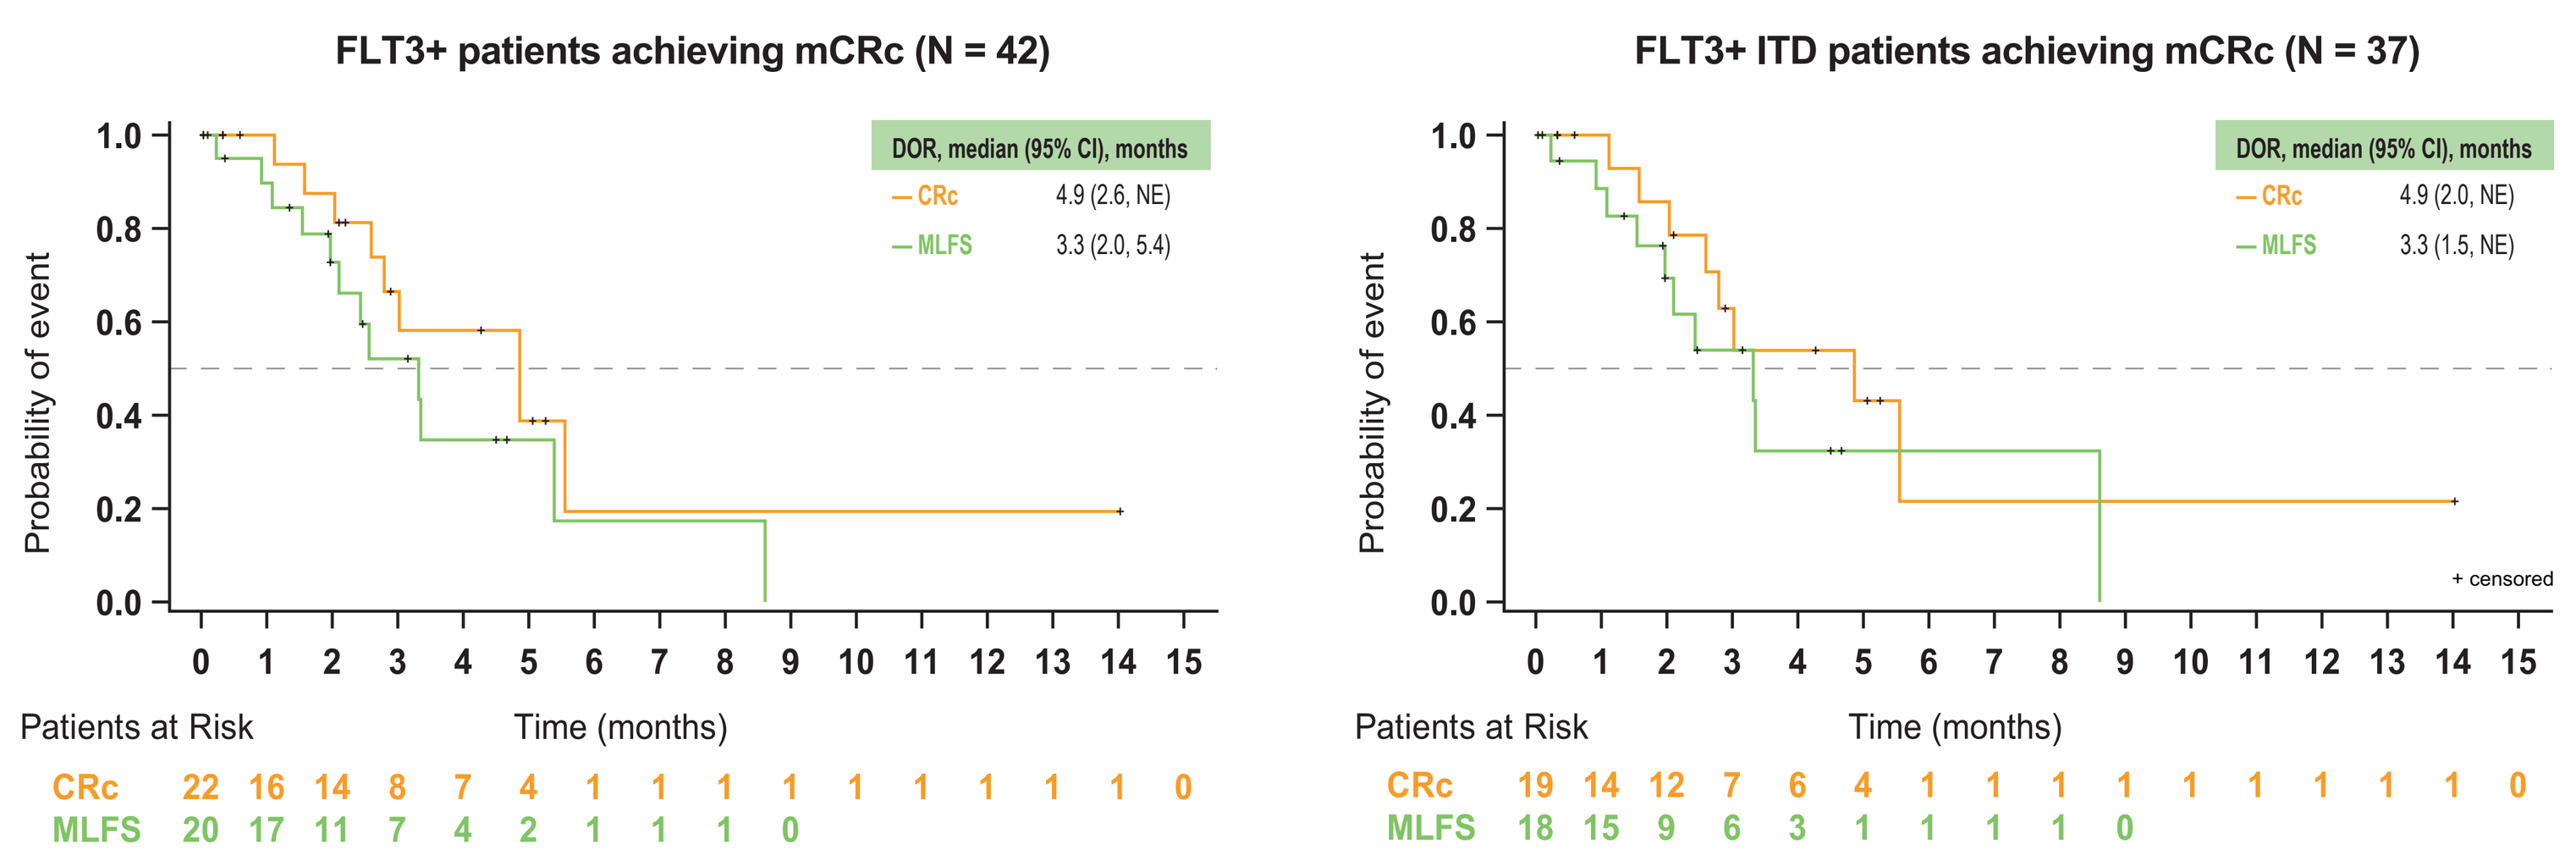
FLT3+ patients achieving mCRc (N = 42)
DOR, median (95% CI), months
CRc 4.9 (2.6, NE)
MLFS 3.3 (2.0, 5.4)
Probability of event
Time (months)
Patients at Risk
CRc 22 16 14 8 7 4 1 1 1 1 1 1 1 1 1 0
MLFS 20 17 11 7 4 2 1 1 1 0
FLT3+ ITD patients achieving mCRc (N = 37)
DOR, median (95% CI), months
CRc 4.9 (2.0, NE)
MLFS 3.3 (1.5, NE)
+ censored
Patients at Risk
CRc 19 14 12 7 6 4 1 1 1 1 1 1 1 1 1 0
MLFS 18 15 9 6 3 1 1 1 1 0

Median (range) duration of Ven: 77 days (2–495)
Median (range) duration of Gilt: 77 days (3–496)

mCRc includes CR/CRp/CRi/MLFS per modified IWG response criteria used in Gilt single agent development.

OVERALL SURVIVAL IN ALL *FLT3*$^{mut+}$ PATIENTS BY TRANSPLANT STATUS AND BEST RESPONSE SUBGROUP

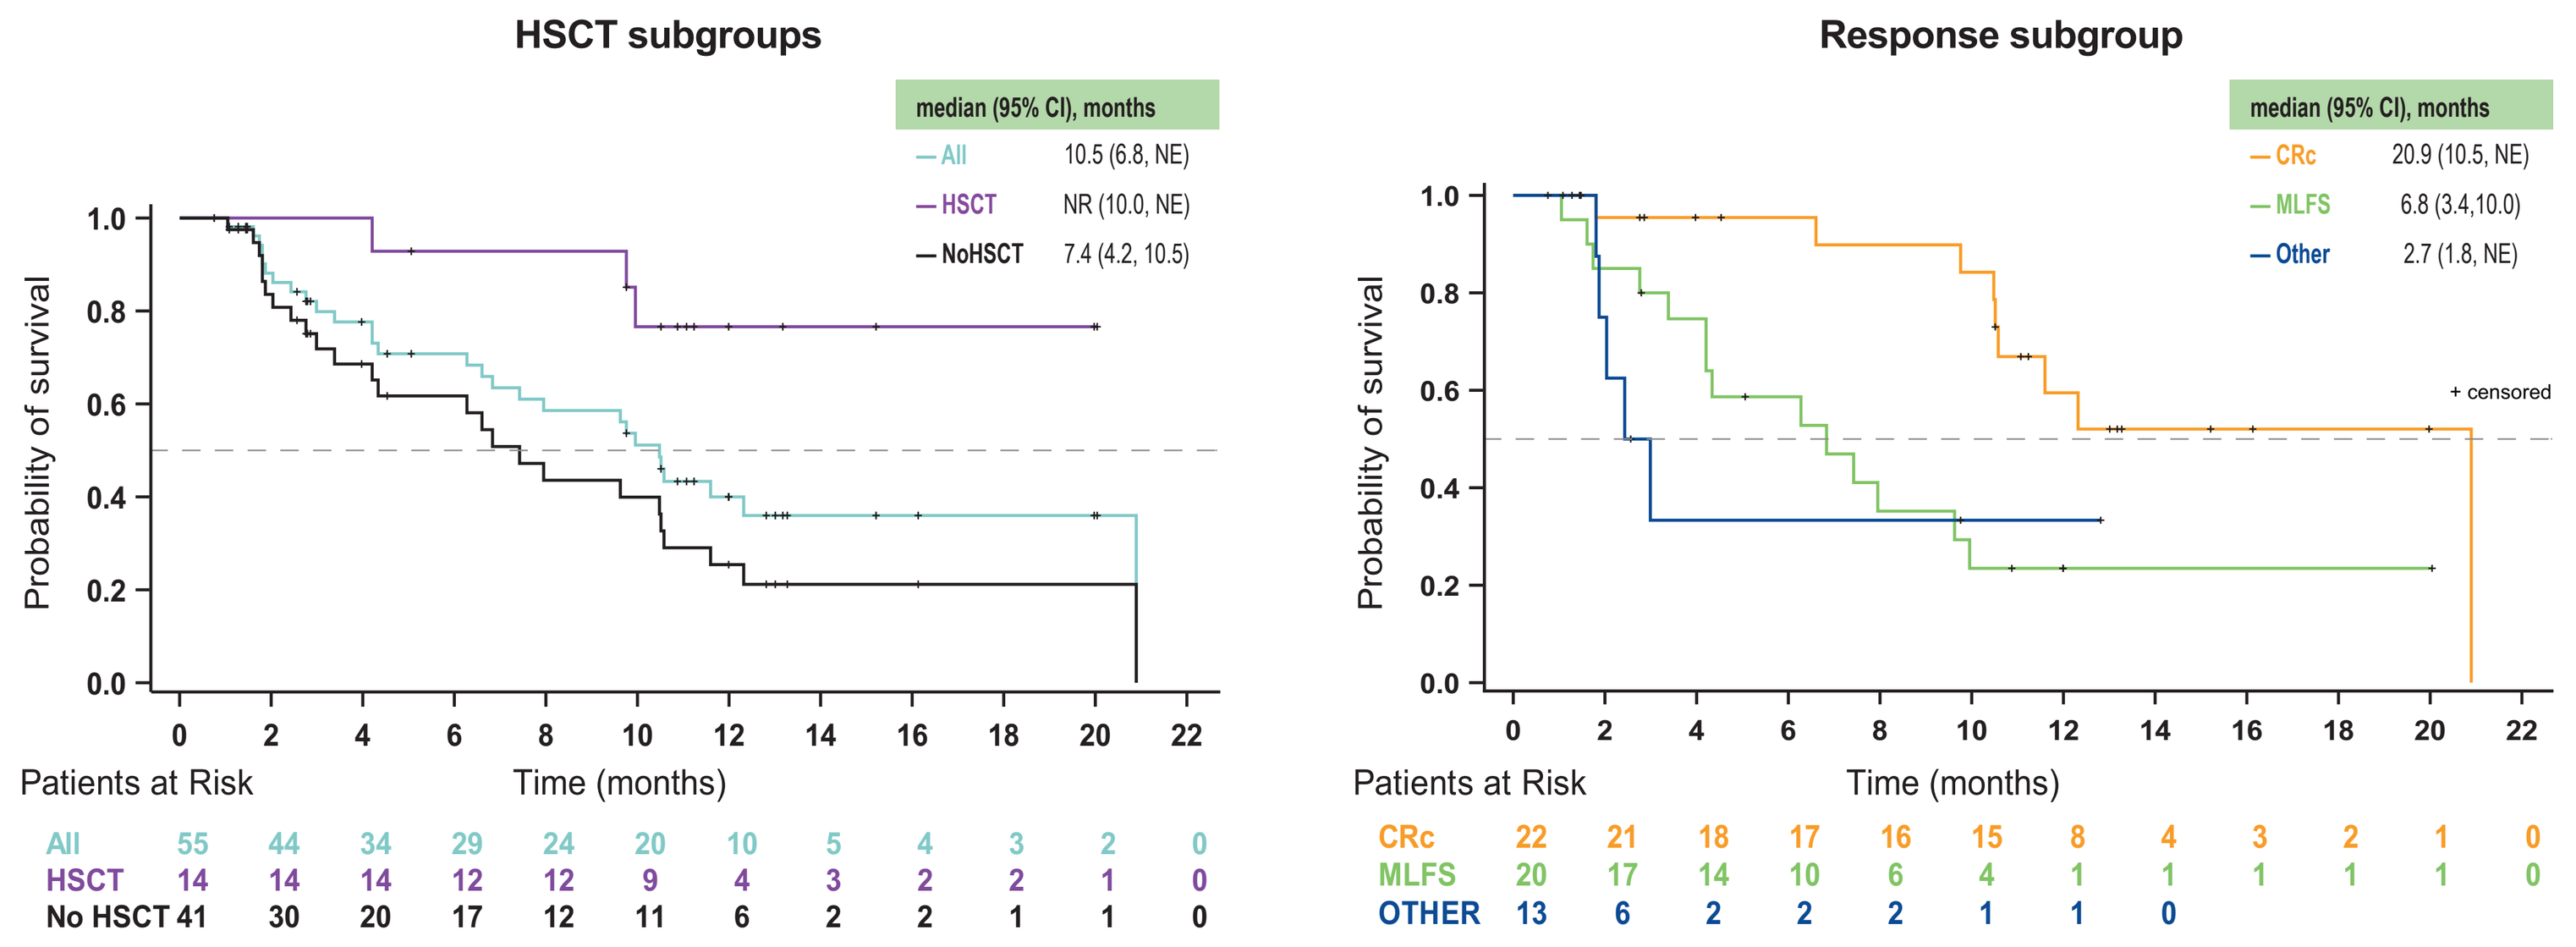
HSCT subgroups
median (95% CI), months
All 10.5 (6.8, NE)
HSCT NR (10.0, NE)
NoHSCT 7.4 (4.2, 10.5)
Probability of survival
Time (months)
Patients at Risk
All 55 44 34 29 24 20 10 5 4 3 2 0
HSCT 14 14 14 12 12 9 4 3 2 2 1 0
No HSCT 41 30 20 17 12 11 6 2 2 1 1 0
Response subgroup
median (95% CI), months
CRc 20.9 (10.5, NE)
MLFS 6.8 (3.4, 10.0)
Other 2.7 (1.8, NE)
+ censored
Patients at Risk
CRc 22 21 18 17 16 15 8 4 3 2 1 0
MLFS 20 17 14 10 6 4 1 1 1 1 1 0
OTHER 13 6 2 2 2 1 1 0

Median (range) duration of follow-up: 12 months (0.8–20.9)

OVERALL SURVIVAL BY PRIOR TKI USE

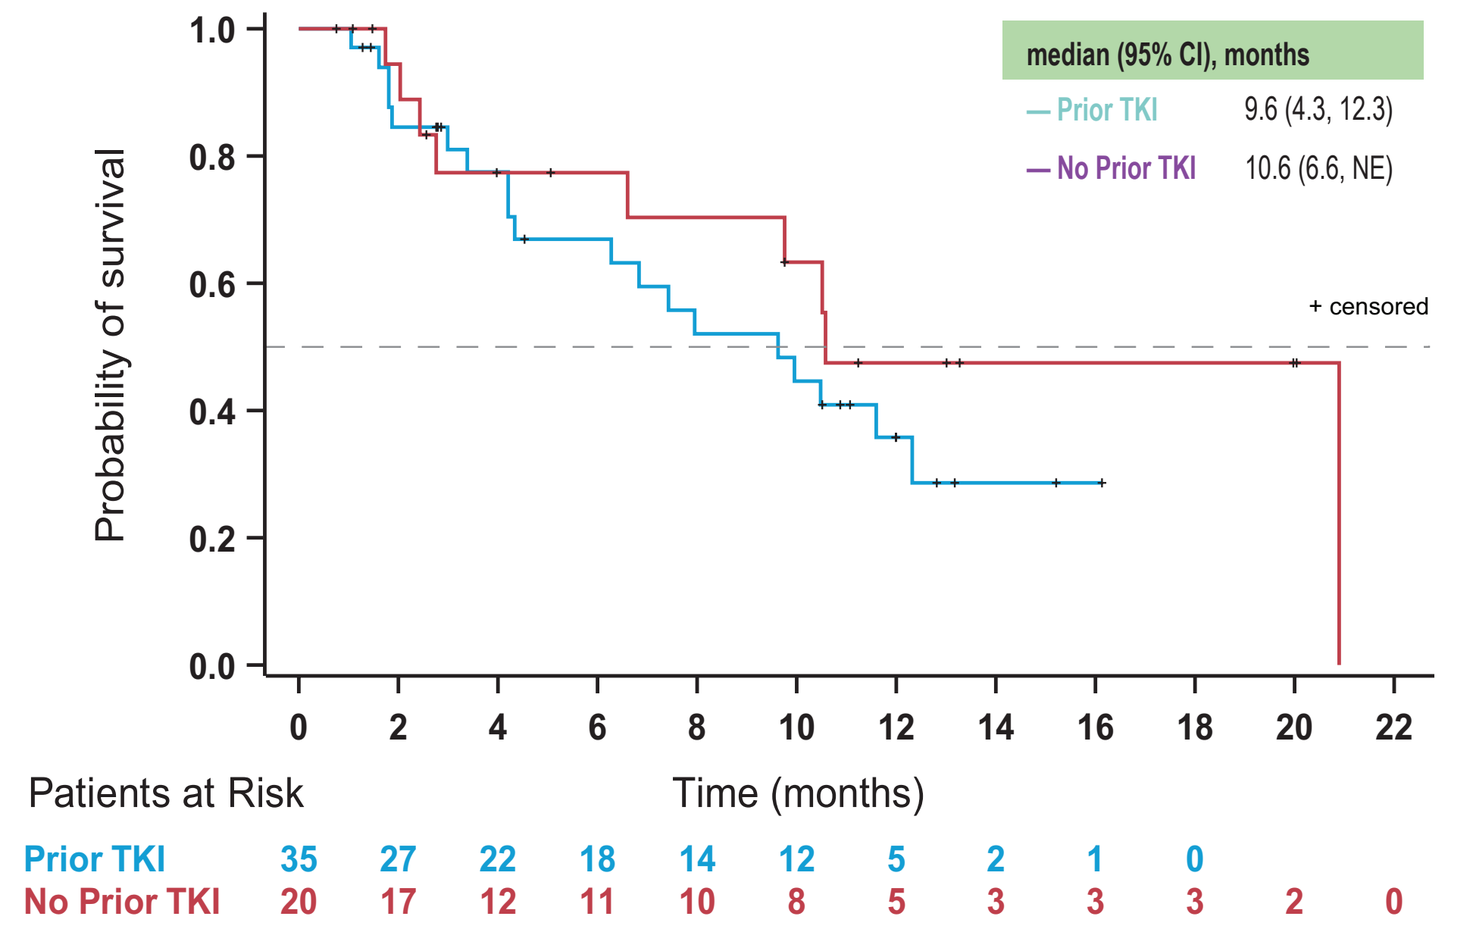
median (95% CI), months
Prior TKI 9.6 (4.3, 12.3)
No Prior TKI 10.6 (6.6, NE)
+ censored
Probability of survival
Time (months)
Patients at Risk
Prior TKI 35 27 22 18 14 12 5 2 1 0
No Prior TKI 20 17 12 11 10 8 5 3 3 3 2 0

VEN + GILT DEMONSTRATED DEEP REDUCTIONS IN FLT3-ITD ALLELIC BURDEN IN PATIENTS ACHIEVING mCRc

- 33/37 FLT3-ITD mCRc patients were evaluable for specific reduction in FLT3-ITD using an assay with sensitivity of 10^{-6}

FLT3-ITD burden, n (%)	$<10^{-2}$ (1%)	$<10^{-3}$	$<10^{-4}$
C1D28	10 (30.3)	3 (9.1)	0
Any time on therapy	20 (60.6)*	14 (42.4)	7 (21.2)

*The molecular best response ($<10^{-2}$) of Ven + Gilt was 60.6%. The molecular best response for Gilt alone in a subset analysis from CHRYSALIS[6] was 25%.

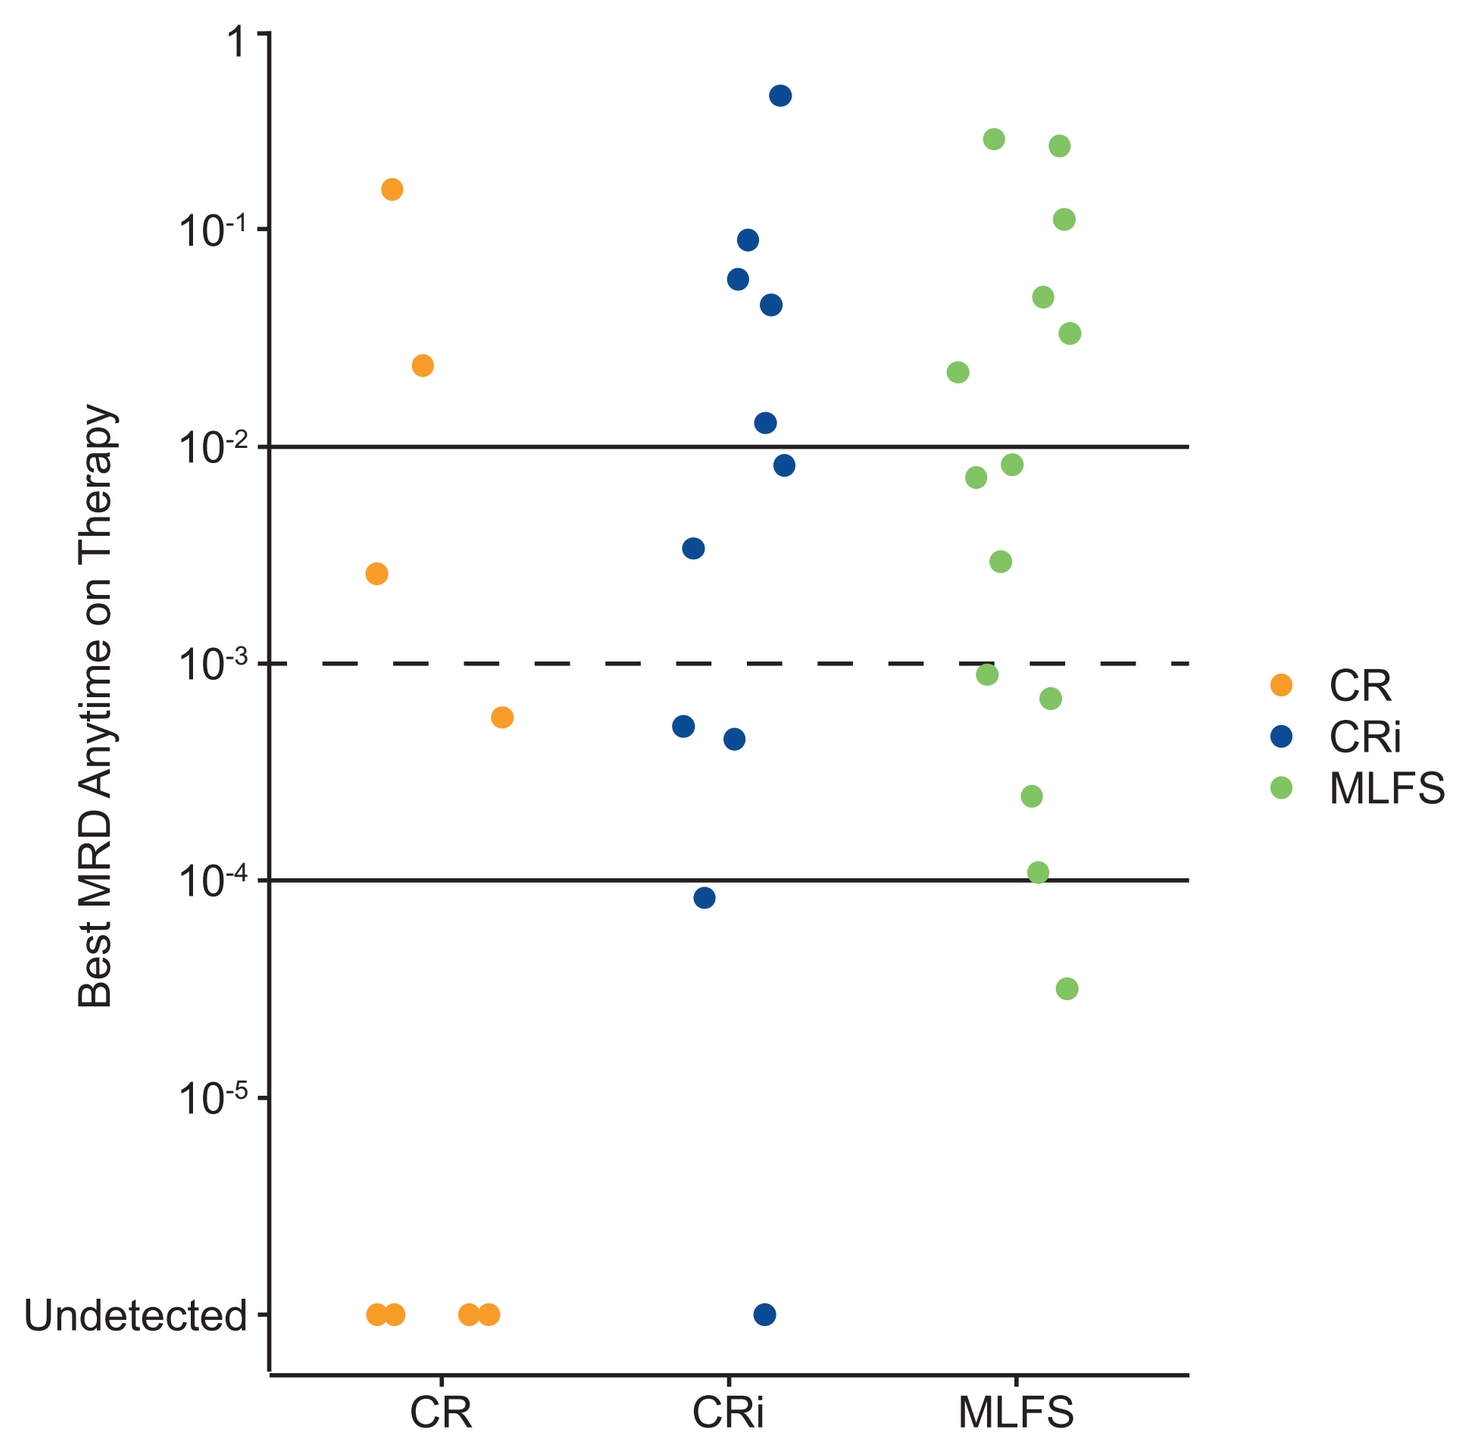
Best MRD Anytime on Therapy
1
10-1
10-2
10-3
10-4
10-5
Undetected
CR
CRi
MLFS

CONCLUSIONS

- Ven + Gilt achieved high rates of mCRc (76.4%) in patients with R/R *FLT3*$^{mut+}$ AML
 - 62.5% of patients had prior FLT3 TKI exposure compared with 12% in ADMIRAL
 - High rates of mCRc (80.0%) was achieved in this group of subjects
- 60.6% of mCRc evaluable patients demonstrated FLT3-ITD reduction
- The overall survival for patients with prior TKI use was 9.6 months and 10.6 months for those with no prior TKI use
 - 30-day mortality rate was 0% and 60-day mortality rate was 10.9%
- Cytopenias were observed but were manageable with appropriate Ven or Gilt dose reductions and/or interruptions
- The updated efficacy and safety results demonstrate the need for continued study of this combination

DISCLOSURES & ACKNOWLEDGMENTS

JK Altman: Research funding to institution: AbbVie, ALX Oncology, Amgen, Aprea, Astellas, BioSight, BMS, Boehringer Ingelheim, Fujifilm, Immunogen, Kartos, Kura. Advisory board: AbbVie, Astellas, Kura Oncology, Syros. Data monitoring committee member: GlycoMimetics. **N Daver:** Research funding: Astellas, AbbVie, Genentech, Daiichi-Sankyo, Gilead, Immunogen, Pfizer, BMS, Trovagene, Servier, Novimmune, Incyte, Hanmi, Fate, Amgen, KITE, Novartis. Advisory/consultancy: Astellas, AbbVie, Genentech, Daiichi-Sankyo, Novartis, Jazz, Amgen, Servier, Karyopharm, Trovagene, Trillium, Syndax, Gilead, Pfizer, BMS, KITE, Actinium. **J Maly:** Nothing to disclose. **M Levis:** Research support: Astellas, FujiFilm. Consulting/honoraria: Astellas, AbbVie, Amgen, BMS, Daiichi-Sankyo, Jazz, Pfizer. **E Ritchie:** Nothing to disclose. **M Litzow:** Research support: AbbVie, Astellas, Amgen, Actinium, Pluristem. Advisory board: Sanofi, NewLink, Omeros. **J McCloskey:** Honoraria and/or consulting fee: Jazz, AbbVie, Takeda, BMS, Amgen. **CC Smith:** Research funding: AbbVie, Revolution Medicines, Fujifilm. Advisory board: Astellas, Daiichi-Sanyko. Honoria: Amgen. **G Schiller:** Research funding: AbbVie, Astellas, Genentech. Speaker honoraria: Astellas. **T Bradley:** Advisory board: AbbVie. Speakers bureau: AbbVie, Novartis. Consulting: Novartis. **RV Tiu:** Employee: Astellas Pharma. Previous employee, stockholder, and patent(s): Eli Lilly and Company. **K Naqvi:** Employee: Genentech and may hold Roche stock or options. **S Siddani, J Wang, P Lee:** Employees: AbbVie and may hold stock or options. **AE Perl:** Research funding: Astellas, AbbVie, Daiichi-Sankyo, Fujifilm. Honoraria and/or consulting fee: Astellas, AbbVie, Arog, BMS/Celgene, Genentech, Daiichi-Sankyo, Actinium, Forma, Syndax, Loxo, Onconova, Sumitomo Dainippon.

Venetoclax is being developed in a collaboration between AbbVie and Genentech, a member of the Roche group. AbbVie, Astellas, and Genentech funded this study and participated in the study design, research, analysis, data collection, interpretation of data, reviewing, and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

The authors, AbbVie, Genentech, Astellas Pharma would like to thank the patients, their families and caregivers, as well as the study investigators, research, and supporting staff. Current AbbVie employees Brenda Chyla and Yan Sun, for MRD analysis and exploratory statistics support. Medical writing support was provided by Rachel M Richardson, an AbbVie employee.

REFERENCES

1. Perl AE, et al. *Lancet Oncol.* 2017;18(8):1061–75.
2. Perl AE, et al. *N Engl J Med.* 2019;381(18):1728–40.
3. Perl AE, et al. *J Clin Oncol.* 2020;38(15):7514.
4. Ma J, et al. *Clin Cancer Res.* 2019;25(22):6815–26.
5. DiNardo C, et al. *N Engl J Med.* 2020;383:617–29.
6. Levis MJ, et al. *Blood Adv.* 2018;2(8):825–31.

Scan QR code to download an electronic version of this poster (MDS 158) and other AbbVie SOHO 2021 Scientific Presentations

QR code expiration: August 8, 2022.

To submit a medical question, please visit www.abbviemedinfo.com.

© AbbVie Inc. 2021