ALL-167

A Phase 1/2 Study to Evaluate the Safety and Efficacy of Ponatinib With Chemotherapy in Pediatric Patients With Relapsed, Resistant, or Intolerant Philadelphia Chromosome–Positive (Ph+) Acute Lymphoblastic Leukemia (ALL) or Who Have the T315I Mutation

Lia Gore,[1] Mignon Loh,[2] Ching-Hon Pui,[3] Michael J Hanley,[4] Jichang Du,[4] Yousif Matloub,[4] Meliessa Hennessy,[4] Muriel Granier,[5] Andrea Biondi,[6] Lewis Silverman[7]

[1]University of Colorado Cancer Center and Children's Hospital, Aurora, CO, USA; [2]UCSF Medical Center-Mission Bay, Helen Diller Family Comprehensive Cancer Center, San Francisco, CA, USA; [3]St. Jude Children's Research Hospital, Memphis, TN, USA; [4]Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited; [5]Incyte Biosciences International Sàrl, Morges, Switzerland; [6]Clinica Pediatrica Universita Milano-Bicocca, Monza, Italy; [7]Dana Farber/Boston's Children's Hospital, Boston, MA, USA

Introduction

Ph+ ALL

- Ph+ ALL accounts for 3% to 5% of pediatric ALL[1,2]
- Current 5-year EFS and OS rates with TKIs + chemotherapy in newly diagnosed pediatric patients with Ph+ ALL are 60% and 86%, respectively.[3] However, the optimal approach for the treatment of pediatric patients with suboptimal responses, resistance, or intolerance to first- and/or second-generation TKIs has not yet been defined

Ponatinib

- Potent third-generation TKI pan-BCR-ABL1 inhibitor that is active against *BCR-ABL1* and all identified single resistance mutations, including the gatekeeper T315I mutation, which confers resistance to other TKIs[4,5]
- Marketing approval in more than 50 countries, including the United States and Europe, in adults with CML or Ph+ ALL that is resistant or intolerant to other TKIs or who have the T3151 mutation[4,5]

Study Aim

- Assess the PK, safety, and tolerability to determine the RP2D of ponatinib (tablet and AAF) and to determine the efficacy as measured by the rate of CR at end of the reinduction block

Study Design

Phase 1/2 | Open label | Single arm | Multicenter: 70 study sites, 16 countries

Phase 1
Daily oral ponatinib + chemotherapy to confirm RP2D for 2 formulations: Tablet (N = 12) and AAF (N = 6)

→ **Phase 2**
Daily oral ponatinib at RP2D + chemotherapy N = 68 evaluable patients (including patients treated at RP2D in Phase 1)

- Phase 1 will establish the RP2D of oral ponatinib in combination with chemotherapy with the chemotherapy backbone; for patients unable to swallow tablets or with a body weight <30 kg, an age-appropriate formulation ([AAF], minitablets) will be used
- A rolling 6 design will be used for both formulation cohorts; additional formulation cohorts may be enrolled at lower or higher doses based on the emerging data

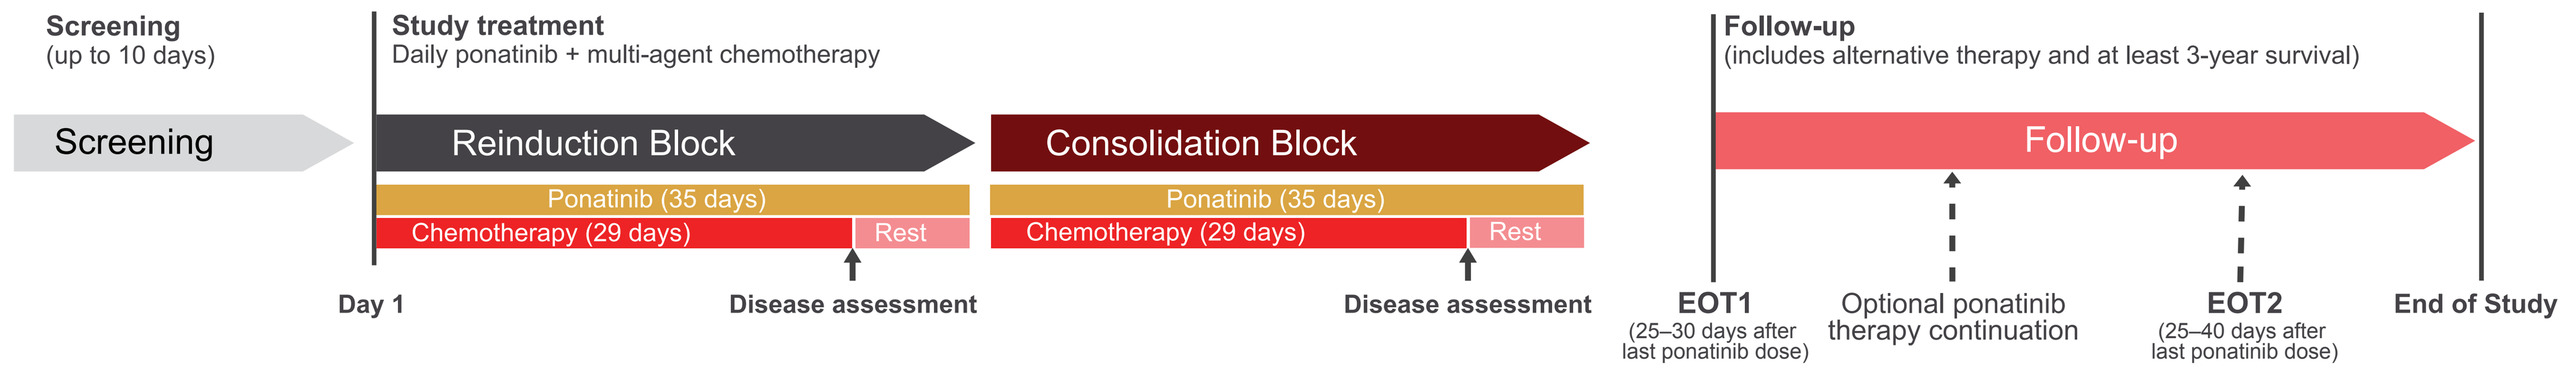
Screening
(up to 10 days)
Study treatment
Daily ponatinib + multi-agent chemotherapy
Follow-up
(includes alternative therapy and at least 3-year survival)
Screening
Reinduction Block
Consolidation Block
Follow-up
Ponatinib (35 days)
Chemotherapy (29 days)
Rest
Ponatinib (35 days)
Chemotherapy (29 days)
Rest
Day 1
Disease assessment
Disease assessment
EOT1
(25–30 days after last ponatinib dose)
Optional ponatinib therapy continuation
EOT2
(25–40 days after last ponatinib dose)
End of Study

Study Treatments

Ponatinib Dosing

	Body Weight			
Oral Ponatinib	**<30 kg**	**30 kg to <45 kg**	**≥45 kg**	**Frequency / Days**
Dose level 1	TBD	20 mg	30 mg	Daily / Days 1–35
Dose level -1 (reduced dosing)		10 mg	15 mg	Daily / Days 1–35

- Ponatinib adult tablet formulation will be used in patients able to swallow tablets
- Each patient's starting dose of ponatinib will be assigned based on body weight
- Initially selected doses are expected to achieve systemic exposures that approximately match adult exposures at 30 mg based on the results of population PK modeling and simulation
- Dose selection for the AAF will be in a separate cohort in phase 1 and informed by the results of a separate relative bioavailability study in healthy adult volunteers[6]

Chemotherapy Regimen: Modified UK ALL R3 Trial[7]

Reinduction Treatment (Weeks 1–4)	Days
Vincristine IV	1, 8, 15, 22
Dexamethasone PO/IV	1–15
PEG-asparaginase IV/IM	2, 15[a]
Daunorubicin IV	1, 2
Initial IT chemotherapy	1
CNS-1/2: IT methotrexate chemotherapy	15, 29
CNS-3: ITT chemotherapy	8, 15, 22, 29

Consolidation Treatment (Weeks 5–8)	Days
Dexamethasone PO/IV	1–5
Vincristine IV	3
Methotrexate IV	8
PEG-asparaginase IV/IM	9
Cyclophosphamide IV	15–19
Etoposide IV	15–19
CNS-1/2 IT methotrexate chemotherapy	8
CNS-3: ITT chemotherapy	8

[a]Day 15 PEG-asparaginase is removed in protocol amendment 2

Inclusion and Exclusion Criteria

Key Inclusion Criteria

- Pediatric population aged ≥1 year to ≤21 years at study entry
- Ph+ status:
 - Ph+ ALL or
 - Ph+ mixed phenotype acute leukemia or
 - Ph-like ALL with targetable kinase-activating lesions (US only)
- Relapsed or resistant/intolerant to ≥1 prior therapy with BCR-ABL1-targeted TKI (if US patient, must have received at least 1 prior second-generation BCR-ABL-targeted TKI)
 Or:
 - *BCR-ABL1* T315I mutation
- Patient weight ≥30 kg (prior to AAF)
- Able to swallow tablets (prior to AAF)
- Performance status:
 - Aged >16 years: Karnofsky PS ≥50%
 - Aged ≤16 years: Lansky Play Scale ≥50%
- Adequate renal and hepatic function with no evidence of pancreatitis

Key Exclusion Criteria

- History or current diagnosis of CML, Burkitt leukemia/lymphoma, or mature B-cell leukemia
- Diagnosis of another concurrent primary malignancy
- Diagnosis of ALL, MPAL, or Ph-like ALL with targetable kinase-activating lesions after cytotoxic therapy for another cancer
- Clinically significant cardiovascular disease, including:
 - Any history of MI or unstable angina
 - History of heart block and/or clinically significant ventricular or atrial arrhythmias
 - Uncontrolled hypertension
- Uncontrolled hypertriglyceridemia (triglycerides ≥450 mg/dL)
- Current systemic use of:
 - Drugs known to prolong QTc intervals or torsades de pointes
 - Drugs/supplements known to be strong inhibitors or inducers of CYP3A within 7 days of the first dose of study drug
- Previous ponatinib treatment
- Known allergies to any study drugs
- Known GI disease/procedures that could interfere with ponatinib absorption

Endpoints

Primary Endpoints

Phase 1: RP2D of ponatinib (tablet and AAF) + chemotherapy

Phase 2: CR rate at the end of the reinduction block with CR defined as[8]:
- No circulating blasts
- <5% blasts in BM
- Normal maturation of cell components in BM
- No extramedullary disease
- ANC >1000/μL (or >1.0 x 10^9/L)
- Platelets >100,000/μL (or >100 x 10^9/L)

Secondary Endpoints

Phase 1
- CR rate at the end of reinduction block
- Characterization of BCR-ABL1 domain mutations prior to and after ponatinib treatment
- Ponatinib PK (C_{max}, T_{max}, AUC_{last})

Phase 2
- Proportion of patients who remain in CR or who achieve CR by the end of consolidation
- Proportion of patients with MRD-negative status (<0.01%) at the end of each block
- Proportion of patients who relapse or progress on study treatment
- Time-to-event estimates EFS, PFS, and OS at 6 months, 1 year, 18 months, 2 years, and 3 years
- Duration of CR
- Proportion of patients who underwent HSCT following study treatment
- Characterization of BCR-ABL1 domain mutations prior to and following ponatinib

Safety Endpoints

Phase 1 & 2
- Adverse events
- Serious adverse events
- Arterial occlusive events
- Venous thromboembolic events
- Adverse events of special interest (eg, pancreatitis)

Exploratory Endpoints

Phase 1 & 2
- Change from baseline in laboratory values and cardiovascular outcomes (elevated cardiac troponin, NT-proBNP)
- Biomarkers of disease sensitivity and resistance to ponatinib and/or biomarkers affecting ponatinib
- Acceptability and palatability of the AAF of ponatinib based on results from Palatability Questionnaire administered during reinduction[9]

Additional Information

Participating sites

- Participating sites will be in the following 16 countries:
 Argentina, Australia, Brazil, China, Czech Republic, France, Hong Kong, Italy, Mexico, The Netherlands, Poland, Portugal, Republic of Korea, Spain, UK, USA

Trial details

- See clinicaltrials.gov
- Clinical trial identifier: NCT04501614

Contact

- Takeda Study Registration Call Center; 1-866-835-2233
- Ask for the ponatinib pediatric trial representative; GlobalOncologyMedinfo@takeda.com

References

1. Aricó M, Schrappe M, Hunger SP, et al. Clinical outcome of children with newly diagnosed Philadelphia chromosome–positive acute lymphoblastic leukemia treated between 1995 and 2005. *J Clin Oncol*. 2010;28(31):4755-4761. **2.** Schlieben S, Borkhardt A, Reinisch I, et al. Incidence and clinical outcome of children with BCR/ABL-positive acute lymphoblastic leukemia (ALL). A prospective RT-PCR study based on 673 patients enrolled in the German pediatric multicenter therapy trials ALL-BFM-90 and CoALL-05-92. *Leukemia*. 1996;10(6):957-963. **3.** Slayton WB, Schultz KR, Kairalla JA, et al. Dasatinib plus intensive chemotherapy in children, adolescents, and young adults with Philadelphia chromosome–positive acute lymphoblastic leukemia: results of Children's Oncology Group Trial AALL0622. *J Clin Oncol*. 2018;36(22):2306-2314. **4.** Cortes JE, Kim DW, Pinilla-Ibarz J, et al. Ponatinib efficacy and safety in Philadelphia chromosome–positive leukemia: final 5-year results of the phase 2 PACE trial. *Blood*. 2018;132(4):393-404. **5.** Zhou T, Commodore L, Huang WS, et al. Structural mechanism of the pan-BCR-ABL inhibitor ponatinib (AP24534): lessons for overcoming kinase inhibitor resistance. *Chem Biol Drug Des*. 2011;77(1):1-11. **6.** Study INCB 84344-103. Relative Bioavailability of Ponatinib MiniTab Capsules and Tablet Administered Orally in Healthy Participants. EudraCT #2018-002335-14. **7.** Parker C, Waters R, Leighton C, et al. Effect of mitoxantrone on outcome of children with first relapse of acute lymphoblastic leukaemia (ALL R3): an open-label randomised trial. *Lancet*. 2010;376(9757):2009-2017. **8.** NCCN Clinical Practice Guidelines in Oncology. Pediatric Acute Lymphoblastic Leukemia. Version 2.2021 — October 22, 2020. **9.** Mulla H, Buck H, Price L, et al. 'Acceptability' of a new oral suspension formulation of mercaptopurine in children with acute lymphoblastic leukaemia. *J Oncol Pharm Pract*. 2016;22(3):387-395.

Abbreviations

AAF, age-appropriate formulation; **ABL**, Abelson; **ALL,** acute lymphoblastic leukemia; **ANC**, absolute neutrophil count; **AUC_{last}**, area under the plasma concentration-time curve from time 0 to the time of the last measurable concentration; **BM**, bone marrow; **C_{max}**, maximum observed concentration; **CML**, chronic myeloid leukemia; **CNS**, central nervous system; **CNS-1/2**, no/very low CNS involvement; **CNS-3**, definite CNS involvement; **CR**, complete remission; **CYP3A**, cytochrome P450 3A; **EFS**, event-free survival; **EOT**, end of treatment; **GI**, gastrointestinal; **HSCT**, hematopoietic stem cell transplantation; **IM**, intramuscular; **IT**, intrathecal; **ITT**, triple intrathecal; **IV**, intravenously; **MI**, myocardial infarction; **MPAL**, mixed phenotype acute leukemia; **MRD**, minimal residual disease; **NT-proBNP**, N-terminal pro-brain natriuretic peptide; **OS**, overall survival; **PEG**, polyethylene glycol; **PFS**, progression-free survival; **Ph+**, Philadelphia chromosome–positive; **Ph-like**, Philadelphia chromosome–like; **PK**, pharmacokinetics; **PO**, orally; **PS**, performance status; **QTc**, corrected QT; **RP2D**, recommended phase 2 dose; **TBD**, to be determined; **TKI**, tyrosine kinase inhibitor; **T_{max}**, time to reach first maximum observed concentration.

Acknowledgments

The authors would like to thank the patients, their families, and their caregivers; the investigators and their team members at each study site; and colleagues from ARIAD Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited.

Professional medical writing assistance was provided Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, USA, and funded by Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited.

Disclosures

The PON-1501 study is sponsored by Takeda Development Center Americas, Inc.

LG: Advisory board, unpaid (Amgen, Novartis, Roche, Kura, OnKure, Pfizer, Syndax); **ML:** Advisory board (Medisix Therapeutics); **C-HP:** Advisory board (Adaptive Biotechnologies) Data Monitoring Committee (Novartis); **MJH:** Employment (Takeda); **JD:** Employment (Takeda); **YM:** Employment (Takeda); **MH:** Employment (Takeda); **MG:** Employment (Incyte); **AB:** Advisory board (Novartis, Celgene, Incyte); **LS:** Advisory board (Takeda, Servier, Syndax, Jazz Pharmaceuticals)

Email address for questions or comments: lia.gore@cuanschutz.edu

For an e-Print, scan this QR code.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors of this poster.

Presented at the Ninth Annual Meeting of the Society of Hematologic Oncology (SOHO 2021), September 8–11, 2021, Houston, TX, USA (Hybrid)