ALL-132

Ponatinib Versus Imatinib With Reduced-Intensity Chemotherapy in Patients With Newly Diagnosed Philadelphia Chromosome–Positive (Ph+) Acute Lymphoblastic Leukemia (ALL): PhALLCON Study

Elias Jabbour,[1] Giovanni Martinelli,[2] Marco Vignetti,[3] Hagop Kantarjian,[1] David Gomez-Almaguer,[4] Yosuke Minami,[5] Herve Dombret,[6] Meliessa Hennessy,[7] Alexander Vorog,[7] Bingxia Wang,[7] Josep-Maria Ribera[8]

[1]The University of Texas, MD Anderson Cancer Center, Houston, TX, USA; [2]IRCCS Istituto Romagnolo per lo Studio dei Tumori "Dino Amadori"- IRST S.r.l., Meldola, Italy; [3]GIMEMA Research Foundation, Rome, Italy; [4]Hospital Universitario, UANL, Monterrey, Nuevo León, Mexico; [5]National Cancer Center Hospital East, Kashiwa, Japan; [6]Institut de Recherche Saint-Louis, Université de Paris, Hôpital Sain-Louis, Paris, France; [7]Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited; [8]Institut Català d'Oncologia-Hospital Germans Trias i Pujol, Josep Carreras Research Institute, Badalona, Barcelona, Spain

Abstract

- **CONTEXT:** Therapies for adults with newly diagnosed Ph+ ALL are limited and associated with poor outcomes. Ponatinib is active against native BCR-ABL1 and all identified single resistance mutations, including T315I. In the PACE study, ponatinib had a 41% major hematologic response rate in heavily pretreated Ph+ ALL, but responses were not durable due to compound mutations. Ponatinib resistance in Ph+ ALL is acquired through compound mutations. Decreased likelihood of compound mutations with first-line ponatinib in Ph+ ALL may lead to more durable responses compared with ponatinib in later lines of therapy. A phase 2 study of ponatinib with chemotherapy in newly diagnosed Ph+ ALL reported improved long-term outcomes vs first/second-generation TKIs.
- **OBJECTIVE:** The PhALLCON trial (NCT03589326) compares efficacy and safety of first-line ponatinib vs imatinib with reduced-intensity chemotherapy.
- **DESIGN**: Phase 3, open-label, parallel-assignment trial. Patients randomized to ponatinib or imatinib with reduced-intensity chemotherapy. Patients remain on study treatment until they complete study, relapse from complete remission (CR), have progressive disease, have unacceptable toxicity, withdraw consent, proceed to hematopoietic stem cell transplant (HSCT), or sponsor terminates study. Study initiated August 2018.
- **SETTING:** As of August 9, 2021, there were 179 patients enrolled; 116 sites and 21 countries
- **PATIENTS:** Trial enrolling 230–320 patients (≥18y) with newly diagnosed Ph+ or BCR-ABL1–positive ALL (p190/p210) and ECOG status ≤2.
- **INTERVENTIONS:** Patients randomized 2:1 to ponatinib 30 mg/d or imatinib 600 mg/d PO with reduced-intensity chemotherapy in induction (cycles [C] 1–3), consolidation (C4–9), and maintenance (C10–20). Ponatinib dose reduced to 15 mg once minimal residual disease–negative (MRD−) CR achieved. After 20C, patients stay on single-agent ponatinib/imatinib. CNS prophylaxis: 2x/mo in C1–6.
- **MAIN OUTCOME MEASURES:** Primary endpoint: MRD− (BCR-ABL/ABL1 ≤0.01%) CR (end of induction). Key secondary: EFS; others include CR/incomplete blood count recovery rates; molecular response rates (MR3, MR4, MR4.5); MRD− CR duration; primary induction failure; overall response rate, CR duration; time to treatment failure; MR4.5 duration after induction; overall survival; and safety, including arterial occlusive/venous thrombotic/embolic events. Subgroup analysis: patients with/without HSCT. Exploratory: mutation status.
- **RESULTS/CONCLUSIONS:** N/A

Introduction

- Therapies for adults with newly diagnosed Ph+ ALL are limited and associated with poor outcomes
- Although the introduction of first- and second-generation BCR-ABL1 tyrosine kinase inhibitors (TKIs) to frontline regimens has proven beneficial, response rates and outcomes remain suboptimal[1-5]
 – A regimen of first- or second-generation TKIs in combination with chemotherapy results in 3- and 5-year overall survival (OS) rates of approximately 56% and 43%, respectively[6-8]
 – Disease progression on first- and second-generation TKIs is highly associated with the development of single resistance mutations in BCR-ABL1, especially T315I (observed in 75% of patients who relapsed on dasatinib)[3]; disease progression in later lines is highly associated with development of resistant compound mutations in BCR-ABL1
- There is a need for a more potent TKI for treating newly diagnosed Ph+ ALL patients that does not lead to the development of single mutations associated with first- and second-generation TKIs
- Ponatinib is a third-generation TKI designed to potently inhibit BCR-ABL1 with or without any single resistance mutation, including T315I
 – In the phase 2 PACE study, Ph+ ALL patients were heavily pretreated (81% had failed ≥2 prior TKIs; 38% had failed ≥3 prior TKIs). Ponatinib had a 41% major hematologic response (MaHR) rate in these patients, but responses were not durable mainly due to compound mutations, with only 8% of Ph+ ALL patients experiencing an estimated sustained MaHR of at least 12 months[9]
 – Resistance to ponatinib in Ph+ ALL is acquired through the development of compound mutations in later lines of treatment[10,11]
- Decreased likelihood of compound mutations with first-line ponatinib in Ph+ ALL may lead to more durable responses compared with ponatinib in later lines of therapy
- Phase 2 trials with newly diagnosed Ph+ ALL patients have shown that ponatinib in combination with steroids or hyper-CVAD is effective in achieving long-term remission[12,13]
 – In patients treated with ponatinib and hyper-CVAD, 3-year event-free survival (EFS) and OS were 70% and 76%, respectively, and 5-year EFS and OS were 67% and 71%, respectively[13]
- The PhALLCON trial described here compares first-line ponatinib vs imatinib with reduced-intensity chemotherapy in newly diagnosed adult Ph+ ALL patients (NCT03589326)

Methods

Figure 1. Study Design

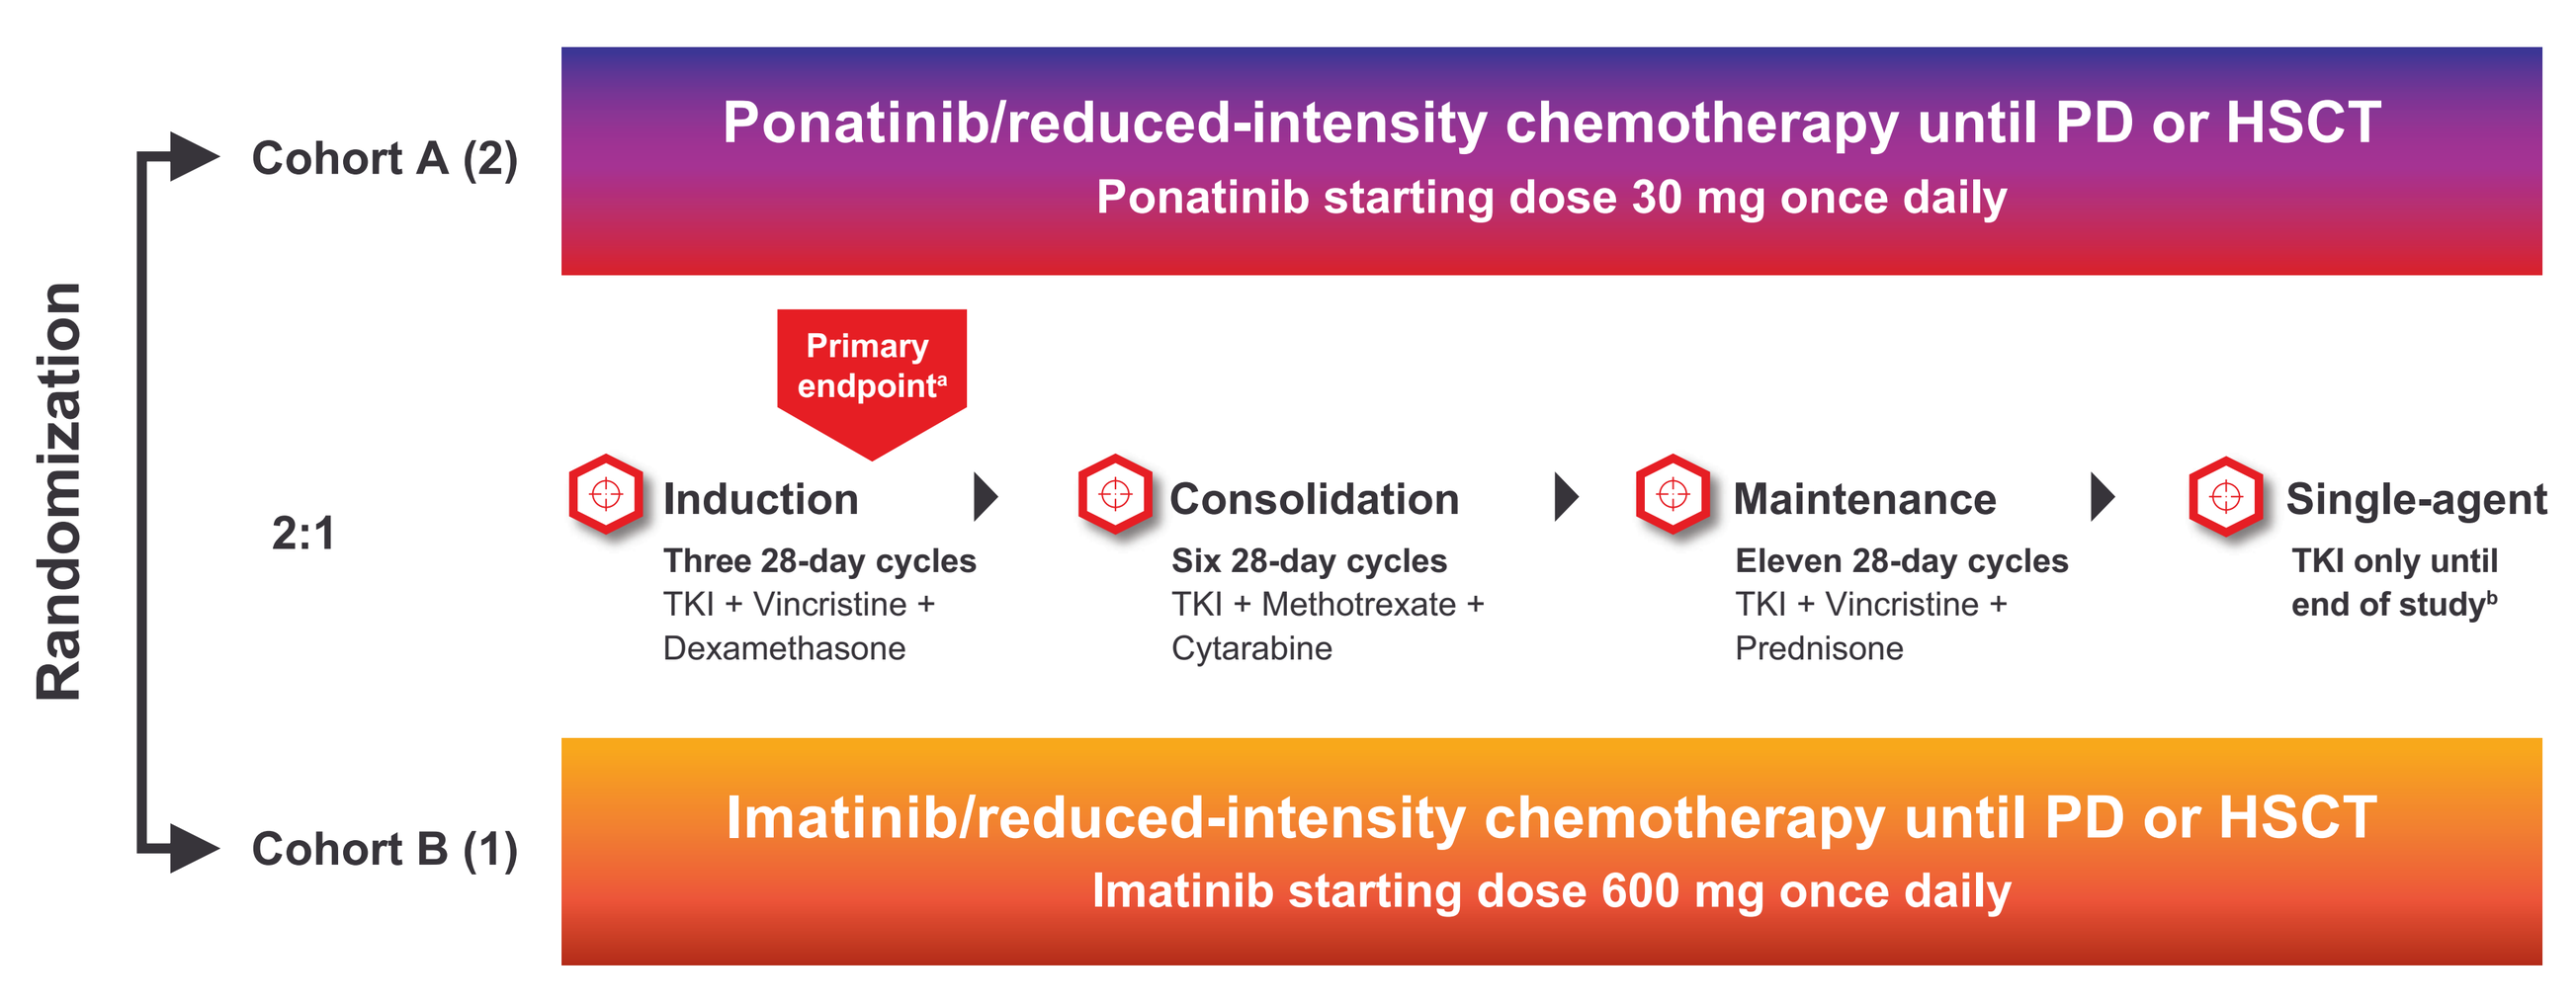

Randomization
2:1
Cohort A (2)
Ponatinib/reduced-intensity chemotherapy until PD or HSCT
Ponatinib starting dose 30 mg once daily
Primary endpoint[a]
Induction
Three 28-day cycles
TKI + Vincristine + Dexamethasone
Consolidation
Six 28-day cycles
TKI + Methotrexate + Cytarabine
Maintenance
Eleven 28-day cycles
TKI + Vincristine + Prednisone
Single-agent
TKI only until end of study[b]
Cohort B (1)
Imatinib/reduced-intensity chemotherapy until PD or HSCT
Imatinib starting dose 600 mg once daily

[a] Primary endpoint: Minimal residual disease–negative (BCR-ABL1/ABL1 ≤0.01%) CR at the end of induction
[b] Following 20 cycles of reduced-intensity chemotherapy regimen, single-agent TKI to be administered continuously until patients have completed the study, experienced relapse from CR or PD, have an unacceptable toxicity, withdraw consent, proceed to HSCT or alternative therapy, or sponsor terminates the study

- **Sample size:** 230 to 320 patients, aged ≥18 years
- **Duration of therapy:** Patients will remain on the study treatment until they complete the study or experience relapse from CR or PD, have unacceptable toxicity, withdraw consent, proceed to HSCT, or until the sponsor terminates the study, whichever occurs first
- **CNS prophylaxis** will be administered (total: 6 cycles, 12 intrathecal injections) and will include a triple intrathecal injection of methotrexate, cytarabine, and corticosteroids per current practice in each center
- Dose modification for safety and efficacy is part of treatment

Figure 2. Study Locations[a]

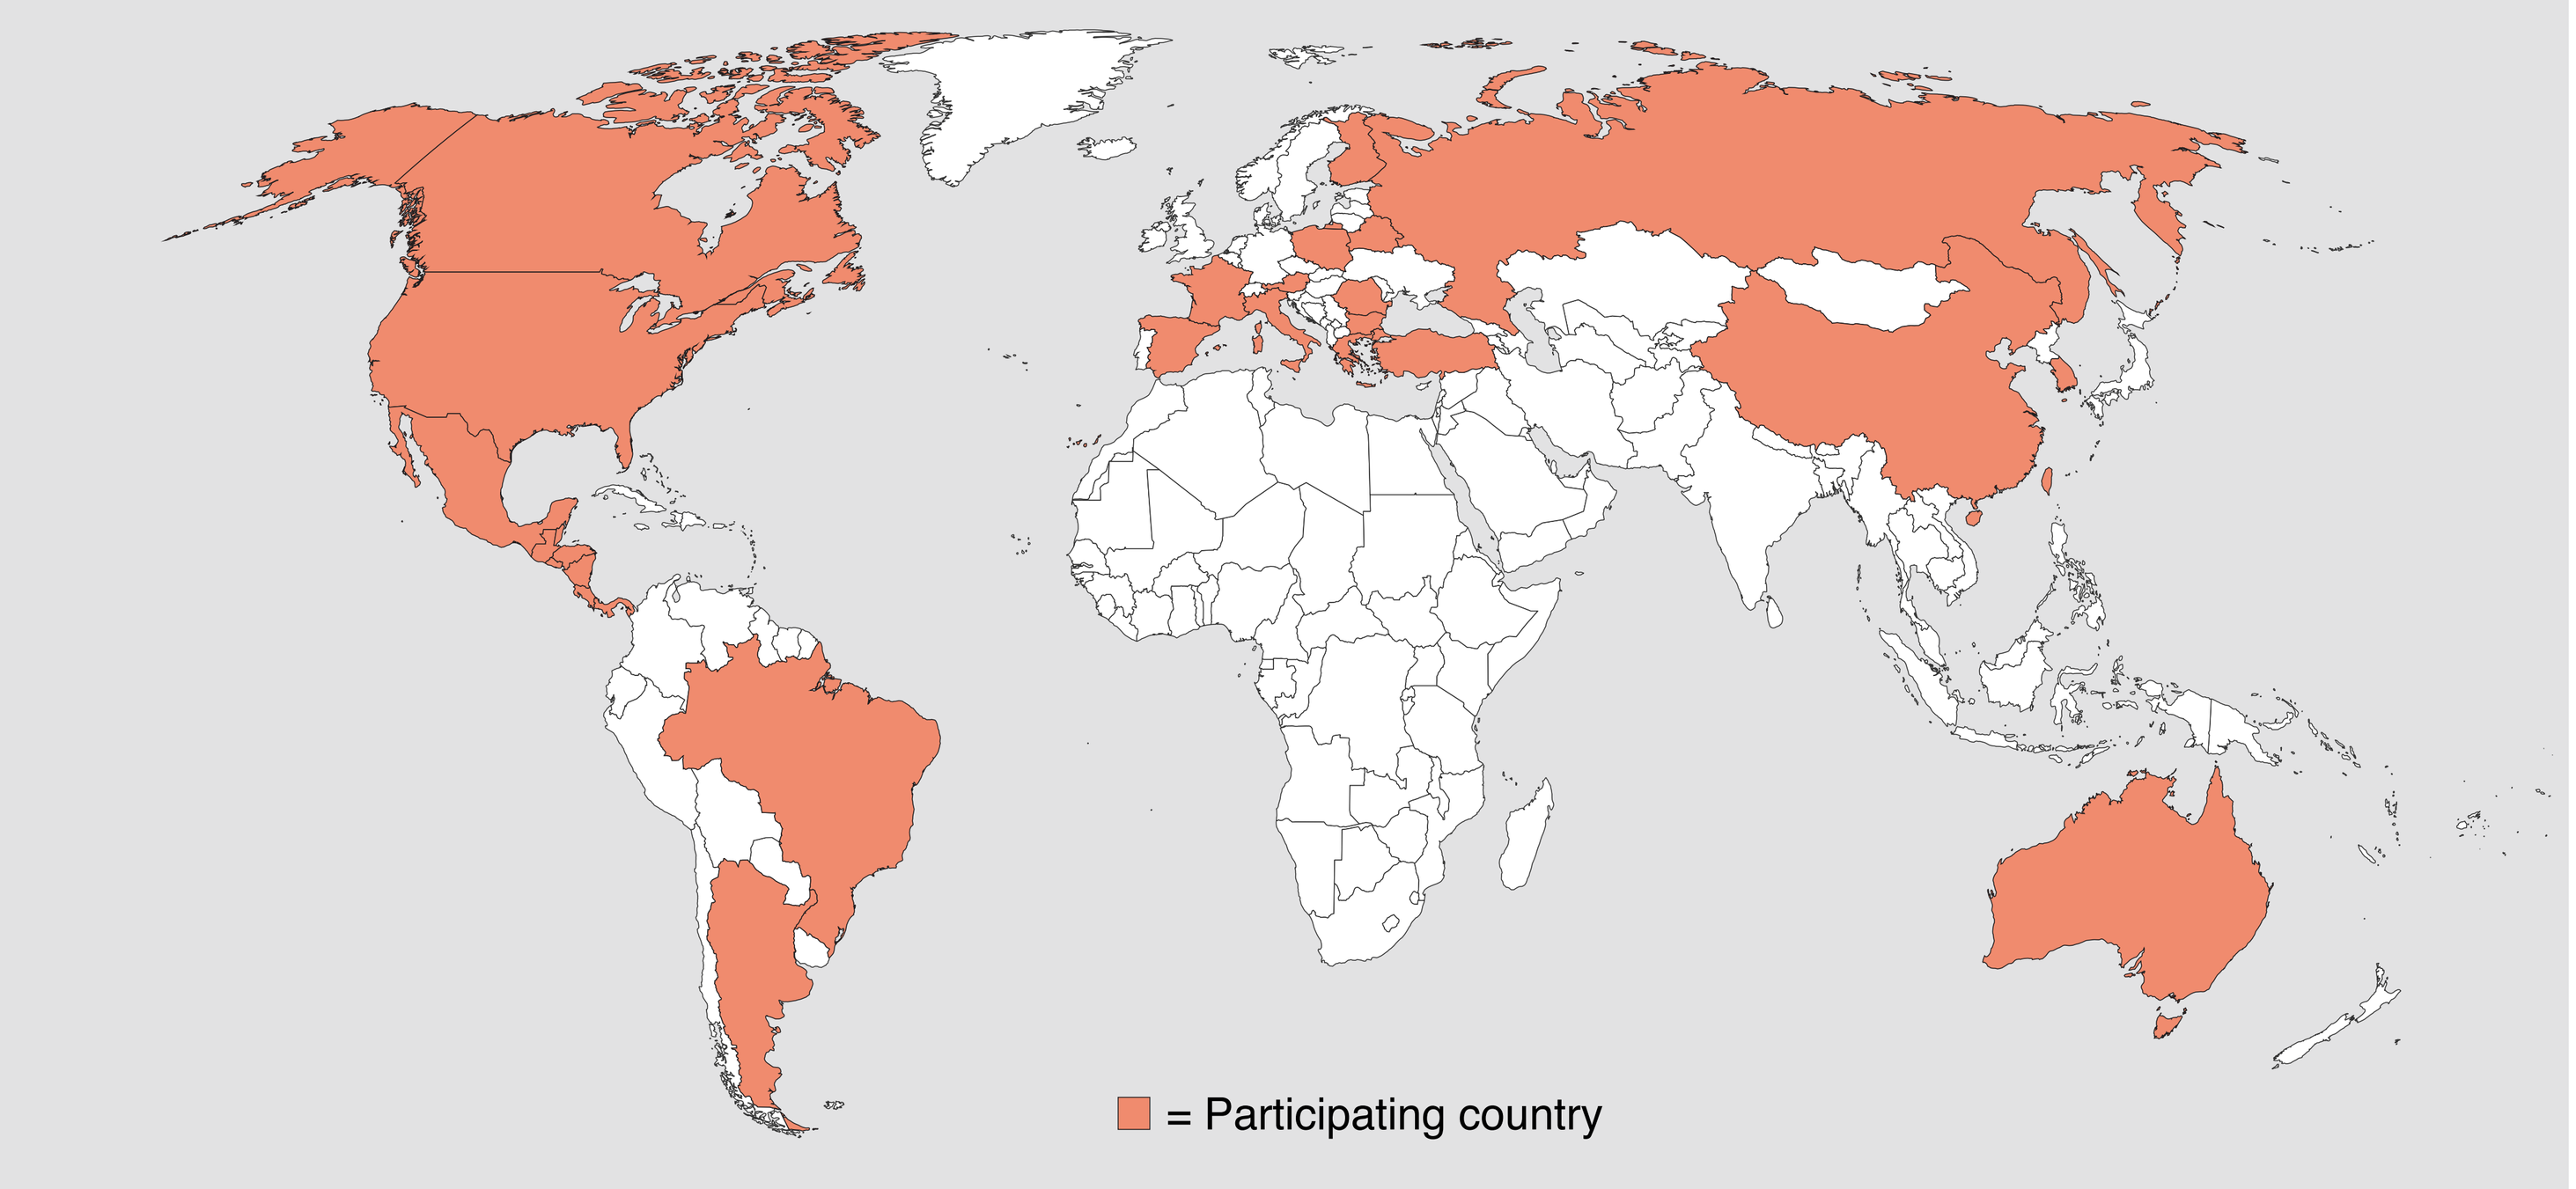

= Participating country

[a] Participating countries: Argentina, Australia, Austria, Belarus, Belize, Brazil, Bulgaria, Canada, China, Costa Rica, El Salvador, Finland, France, Greece, Guatemala, Honduras, Italy, Mexico, Nicaragua, Panama, Poland, Romania, Russia, South Korea, Spain, Taiwan, Turkey, and the United States

- Approximately 320 patients will be enrolled at ≈115 sites across 21 countries in North, Central, and South America, Europe, and the Asia-Pacific region
- Trial opened for enrollment in August 2018
- Accrual is ongoing with 179 patients currently randomized (as of August 9, 2021)

Table 1. Eligibility Criteria

Main Inclusion Criteria

- ≥18 years of age
- Newly diagnosed Ph+ or BCR-ABL1–positive ALL, as defined by the 2017 NCCN Guidelines
- Molecular assessment of BCR-ABL1 must demonstrate the presence of a p190 (ie, e1a2) or p210 (ie, e13a2 or e14a2 [also known as b2a2 or b3a2]) transcript type
- ECOG performance status ≤2
- Normal organ function within 30 days before randomization
- Normal QT interval corrected per Fridericia method (QTcF) on screening electrocardiogram, defined as QTcF of ≤450 ms in males or ≤470 ms in females

Main Exclusion Criteria

- History or current diagnosis of chronic, accelerated, or blast phase CML
- Prior/current treatment with any systemic anticancer therapy and/or radiotherapy for ALL, with the exception of an optional prephase therapy or chemotherapy induction (no more than 1 cycle)
- Currently taking medications known to have risk of causing prolonged QTc or torsades de pointes
- Taking medications or herbal supplements known to be strong inhibitors or inducers of CYP3A4 within at least 14 days before first dose of study drug
- Uncontrolled active serious infections that could, in the investigator's opinion, potentially interfere with the completion of treatment according to this protocol
- Major surgery within 28 days before randomization
- History of acute pancreatitis within 1 year of study screening or history of chronic pancreatitis
- Uncontrolled hypertriglyceridemia (triglycerides >450 mg/dL)
- Primary malignancy other than ALL within 5 years before randomization or previously diagnosed with another malignancy with evidence of residual disease. Nonmelanoma skin cancer and carcinoma in situ are not excluded if they have undergone complete resection
- History or presence of clinically relevant CNS pathology or autoimmune disease with potential CNS involvement
- Clinical manifestations of CNS or extramedullary involvement with ALL
- Clinically significant, uncontrolled, or active cardiovascular, cerebrovascular, or peripheral vascular disease, or history of or active VTE disease
- Known significant neuropathy grade ≥2 severity

Endpoints

- Primary endpoint: Minimal residual disease–negative (BCR-ABL1/ABL1 ≤0.01%) CR at the end of induction
- Secondary endpoints:
 – Event-free survival
 – CR and incomplete blood count recovery rates at the end of cycles 1 and 2, end of induction, and during or end of consolidation
 – Molecular response rates (MR3, MR4, MR4.5) at the end of cycles 1 and 2, end of induction, and during or end of consolidation
 – Primary induction failure rate at the end of induction
 – ORR rate at the end of induction
 – Minimal residual disease–negative CR rates at multiple intervals after the end of induction
 – Duration of minimal residual disease–negative CR
 – Duration of CR
 – Time to treatment failure
 – MR4.5 rates (including best response) at multiple intervals including end of induction, end of consolidation, and prior to HSCT
 – Duration of MR4.5 in patients who achieve MR4.5
 – OS
 – Safety, including arterial occlusive/venous thrombotic/embolic events
- Subgroup analysis: Patients with or without HSCT, including rates of OS and relapse from CR
- Exploratory endpoints include quality of life and biomarkers of disease sensitivity and resistance to ponatinib and imatinib

Figure 3. Statistical Methods

Primary analysis of the primary endpoint

- Analysis will be based on the ITT population (all patients who are randomized)
- Analysis will test for differences comparing the proportion of patients who achieve the primary endpoint at the end of induction in the ponatinib vs imatinib arms
- The O'Brien-Fleming alpha spending function (Lan-DeMets method) will be used to calculate significance and futility boundaries at IA and FA. With 65% information available at IA, the efficacy boundary will be:
 – 0.011 at IA, conducted using CMH chi-square test; significance level designated by the alpha spending assigned by the group sequential procedure, with stratification according to the stratification factors
 – 0.046 at FA, conducted using the Cui-Hung-Wang test, a weighted statistic based on CMH test results at IA and FA

Interim analysis

- One IA is planned and will be performed after the end-of-induction–phase data have been collected for 150 patients
- If the 2-sided *P* value is greater than the alpha spending cutoff at IA, or if the enrollment is not terminated after IA for futility, the conditional power for the primary endpoint will be calculated
- If the conditional power falls in the:
 – Favorable or unfavorable zones, the preplanned sample size for FA of minimal residual disease–negative (BCR-ABL1/ABL1 ≤0.01%) CR at the end of induction, CR will remain at 230 patients
 – Promising zone, the sample size will be determined according to a prespecified sample size adaptation rule, with a cap of approximately 320 patients

Rationale for number of patients

- Conditional power-based sample size re-estimation adaptive design is used in determining sample size and trial power[14,15]
- Assuming an optimistic effect size of 28% (48% and 20% minimal residual disease–negative [BCR-ABL1/ABL1 ≤0.01%] CR rates at the end of induction for the active and control arms, respectively), an upfront committed sample size of 230 patients (ponatinib, n=153; imatinib, n=77, based on a 2:1 allocation ratio) will provide 83% power at IA
- If enrollment is not terminated after IA for futility, conditional power will be calculated
 – If conditional power falls in the favorable or unfavorable zones, sample size for FA will remain at 230 patients
 – If conditional power falls in the promising zone, sample size will be determined according to a prespecified sample size adaptation rule, with a cap of approximately 320 patients providing an overall study power of 82.6% if the actual effect size is 17% (40% and 23% minimal residual disease–negative [BCR-ABL1/ABL1 ≤0.01%] CR rates at the end of induction for ponatinib and imatinib arms, respectively)
- Sample size adaptation rule is a prespecified stepwise function to avoid the possibility of back calculation of the trial IA outcome, as 1 sample size recommendation will correspond to multiple IA results, either barely promising or highly promising

Summary

- This trial is evaluating first-line ponatinib vs imatinib with reduced-intensity chemotherapy in patients with newly diagnosed Ph+ or BCR-ABL1–positive ALL
- The primary endpoint is minimal residual disease–negative CR (CR and BCR-ABL1/ABL1 ≤0.01%) at the end of induction
- Planned enrollment is 230–320 patients
- Accrual is ongoing in 21 countries across North, Central, and South America, Europe, and the Asia-Pacific region

References

1. Fielding AK, Rowe JM, Buck G, et al. UKALLXII/ECOG2993: addition of imatinib to a standard treatment regimen enhances long-term outcomes in Philadelphia positive acute lymphoblastic leukemia. *Blood*. 2014;123:843-850. **2.** Vignetti M, Fazi P, Cimino G, et al. Imatinib plus steroids induces complete remissions and prolonged survival in elderly Philadelphia chromosome–positive patients with acute lymphoblastic leukemia without additional chemotherapy: results of the Gruppo Italiano Malattie Ematologiche dell'Adulto (GIMEMA) LAL0201-B protocol. *Blood*. 2007;109:3676-3678. **3.** Rousselot P, Coudé MM, Gokbuget N, et al; European Working Group on Adult ALL (EWALL) group. Dasatinib and low-intensity chemotherapy in elderly patients with Philadelphia chromosome–positive ALL. *Blood*. 2016;128:774-782. **4.** Kim D-Y, Joo Y-D, Lim S-N, et al; Adult Acute Lymphoblastic Leukemia Working Party of the Korean Society of Hematology. Nilotinib combined with multiagent chemotherapy for newly diagnosed Philadelphia-positive acute lymphoblastic leukemia. *Blood*. 2015;126:746-756. **5.** Jabbour E, DerSarkissian M, Duh MS, et al. Efficacy of ponatinib versus earlier generation tyrosine kinase inhibitors for front-line treatment of newly diagnosed Philadelphia-positive acute lymphoblastic leukemia. *Clin Lymphoma Myeloma Leuk*. 2018;18:257-265. **6.** Jabbour E, Kantarjian H, Ravandi F, et al. Combination of hyper-CVAD with ponatinib as first-line therapy for patients with Philadelphia chromosome-positive acute lymphoblastic leukaemia: a single-centre, phase 2 study. *Lancet Oncol*. 2015;16:1547-1555. **7.** Sasaki K, Jabbour EJ, Ravandi F, et al. Hyper-CVAD plus ponatinib versus hyper-CVAD plus dasatinib as frontline therapy for patients with Philadelphia chromosome–positive acute lymphoblastic leukemia: a propensity score analysis. *Cancer*. 2016;122:3650-3656. **8.** Daver N, Thomas D, Ravandi F, et al. Final report of a phase II study of imatinib mesylate with hyper-CVAD for the front-line treatment of adult patients with Philadelphia chromosome–positive (Ph+) acute lymphoblastic leukemia. *Haematologica*. 2015;100:653-661. **9.** Cortes JE, Kim D-W, Pinilla-Ibarz J, et al. Ponatinib efficacy and safety in Philadelphia chromosome–positive leukemia: final 5-year results of the phase 2 PACE trial. *Blood*. 2018;132:393-404. **10.** O'Hare T, Shakespeare WC, Zhu X, et al. AP24534, a pan-BCR-ABL inhibitor for chronic myeloid leukemia, potently inhibits the T315I mutant and overcomes mutation-based resistance. *Cancer Cell*. 2009;16:401-412. **11.** Zabriskie MS, Eide CA, Tantravahi SK, et al. BCR-ABL1 compound mutations combining key kinase domain positions confer clinical resistance to ponatinib in Ph chromosome-positive leukemia. *Cancer Cell*. 2014;26:428-442. **12.** Martinelli G, Piciocchi A, Papayannidis C, et al. First report of the Gimema LAL1811 phase II prospective study of the combination of steroids with ponatinib as frontline therapy of elderly or unfit patients with Philadelphia chromosome-positive acute lymphoblastic leukemia. *Blood*. 2017;130:99. **13.** Jabbour E, Short NJ, Ravandi F, et al. Combination of hyper-CVAD with ponatinib as first-line therapy for patients with Philadelphia chromosome-positive acute lymphoblastic leukaemia: long-term follow-up of a single-centre, phase 2 study. *Lancet Haematol*. 2018;5:e618-627. **14.** Mehta CR, Pocock SJ. Adaptive increase in sample size when interim results are promising: a practical guide with examples. *Stat Med*. 2011;30:3267-3284. **15.** Hung HM, Wang SJ, O'Neill RT. Methodological issues with adaptation of clinical trial design. *Pharm Stat*. 2006;5:99-107.

Abbreviations

ALL, acute lymphoblastic leukemia; BCR-ABL, breakpoint cluster region-Abelson; CMH, Cochran-Mantel-Haenszel; CML, chronic myeloid leukemia; CNS, central nervous system; CR, complete remission (complete response); ECOG, Eastern Cooperative Oncology Group; EFS, event-free survival; FA, final analysis; HbA1c, glycosylated hemoglobin; HSCT, hematopoietic stem cell transplantation; hyper-CVAD, hyper-fractionated cyclophosphamide, vincristine, doxorubicin, and dexamethasone; IA, interim analysis; ITT, intent-to-treat; MaHR, major hematologic response; MR3, molecular response 3-log reduction (BCR-ABL1/ABL1 ≤0.1%); MR4, molecular response 4-log reduction (BCR-ABL1/ABL1 ≤0.01%); MR4.5, molecular response 4.5-log reduction (BCR-ABL1/ABL1 ≤0.0032%); MRD–, minimal residual disease–negative; NCCN, National Comprehensive Cancer Network; ORR, overall response rate; OS, overall survival; PACE, Ponatinib Ph+ ALL and CML Evaluation; PD, progressive disease; Ph+, Philadelphia chromosome–positive; QTcF, QT interval corrected per Fridericia method; TKI, tyrosine kinase inhibitor; VTE, venous thromboembolism

Acknowledgments

The authors would like to thank the patients, their families, and their caregivers; the study investigators and their team members at each study site; and colleagues from Millennium Pharmaceuticals, Inc., Cambridge, MA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited. Professional medical writing assistance was provided by Lauren Gallagher, RPh, PhD, of Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, USA, and funded by Millennium Pharmaceuticals, Inc., a wholly owned subsidiary of Takeda Pharmaceutical Company Limited.

Disclosures

This study is sponsored by Millennium Pharmaceuticals, Inc., Cambridge, MA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited.

EJ: Research grants and advisory roles (AbbVie, Adaptive Biotechnologies, Amgen, BMS, Genentech, Pfizer, Takeda); GM: Speaker and advisory board/consultancy (Astellas, Takeda, Amgen, Incyte, Pfizer, Roche, Jazz Pharmaceuticals, AbbVie, Menarini), speakers bureau (Novartis, Janssen, Celgene), research support/funding (all to institution: Pfizer, Daiichi Sankyo, Incyte); MV: Speakers bureau (Jazz Healthcare Italy SRL, Amgen, Millennium Pharmaceuticals, Celgene, Janssen, Novartis, Incyte); HK: Honoraria (AbbVie, Amgen, ARIAD, BMS, Immunogen, Orsenix, Pfizer, Agios, Takeda, Actinium Pharmaceuticals), research funding (all to institution: Pfizer, Amgen, BMS, Novartis, ARIAD, Astex Pharmaceuticals, AbbVie, Agios, Cyclacel, Immunogen, Jazz Pharmaceuticals); DGA: Speaker and advisory board (Takeda, Amgen, Novartis, BMS, AbbVie, Janssen); YM: Honoraria and advisory roles (Novartis, BMS, Pfizer, Takeda); HD: Honoraria (Celgene, Pfizer, Novartis, Jazz Pharmaceuticals, Amgen, Daiichi Sankyo, Incyte, Astellas, Menarini, AbbVie, Otsuka), research funding (all to institution: Amgen, Pfizer, Novartis, Servier, Jazz Pharmaceuticals, Astellas); MH: Employment (Takeda); AV: Employment (Takeda); BW: Employment (Takeda); JMR: Speaker and advisory boards (Pfizer, Amgen, Shire, Incyte), clinical trials (Pfizer, Amgen, Incyte), research support (Amgen).

Email address for questions or comments: ejabbour@mdanderson.org

For an e-Print, scan this QR code.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors of this poster.

Presented at the Ninth Annual Meeting of the Society of Hematologic Oncology (SOHO 2021), September 8–11, 2021, Houston, TX, USA (Hybrid)