Overall Survival with Tafasitamab + Lenalidomide versus Routinely Administered Therapies for ASCT-Ineligible Relapsed or Refractory Diffuse Large B-Cell Lymphoma: Outcomes from the Observational RE-MIND2 Study

Grzegorz S. Nowakowski,[1] Dok Hyun Yoon,[2] Patrizia Mondello,[3] Erel Joffe,[3] Anthea Peters,[4] Isabelle Fleury,[5] Richard Greil,[6] Matthew Ku,[7] Reinhard Marks,[8] Kibum Kim,[9] Pier Luigi Zinzani,[10] Judith Trotman,[11] Dan Huang,[12] Eva E. Waltl,[12] Mark Winderlich,[12] Sumeet Ambarkhane,[12] Georg Hess,[13] Gilles Salles[3]

[1]Division of Hematology, Mayo Clinic, Rochester, MN, USA; [2]Department of Oncology, Asan Medical Center, Songpa-gu, Seoul, South Korea; [3]Department of Medicine, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [4]Department of Oncology, University of Alberta, Edmonton, Alberta, Canada; [5]Maisonneuve-Rosemont Hospital, Institut Universitaire d'Hémato-Oncologie-Transplantation Cellulaire, Montreal University, Canada; [6]Paracelsus Medical University Salzburg, Salzburg Cancer Research Institute-CCCIT, and Cancer Cluster Salzburg, Austria; [7]Department of Haematology, St Vincent's Hospital and University of Melbourne, Melbourne, Victoria, Australia; [8]University Hospital Freiburg Internal Medicine I, Freiburg im Breisgau, Germany; [9]University of Utah, Salt Lake City, UT, USA; [10]IRCCS Azienda Ospedaliero-Universitaria di Bologna Istituto di Ematologia "Seràgnoli" & Dipartimento di Medicina Specialistica, Diagnostica e Sperimentale Università di Bologna, Bologna, Italy; [11]Haematology Department, Concord Repatriation General Hospital, University of Sydney, Concord, NSW, Australia; [12]MorphoSys AG, Planegg, Germany; [13]Department of Hematology, Oncology and Pneumology, University Medical Center, Johannes Gutenberg-University Mainz, Germany

SOHO September 8–11, 2021: Poster number ABCL–346

Background

- Diffuse large-B cell lymphoma (DLBCL) is the most common subtype of non-Hodgkin's lymphoma, accounting for up to 45% of new lymphoma cases reported annually[1]
- The National Comprehensive Cancer Network® (NCCN) guidelines recommend R-CHOP (rituximab, cyclophosphamide, doxorubicin, vincristine, and prednisone) as first-line treatment for patients with newly diagnosed DLBCL[2]
 - Cure rates with R-CHOP range from 60 to 70%, with ~40% of patients experiencing primary refractory disease or relapse after initial response[3,4]
- Salvage therapy for patients with relapsed/refractory (R/R) disease comprises chemotherapy followed by high-dose chemotherapy,[2] and autologous stem cell transplant (ASCT) is associated with relapse in 30–60% of patients[5–7]
- Second-line treatment options for ASCT-ineligible patients (e.g. due to advanced age and comorbidities) include chemoimmunotherapy regimens, such as bendamustine + rituximab (BR), and rituximab + gemcitabine and oxaliplatin (R-GemOx)[2]
 - Median overall survival (OS) is reported as 4.7 months with BR[8] and 10 months with R-GemOx[9]
- In the single-arm, Phase II L-MIND study (NCT02399085), the chemotherapy-free tafasitamab + lenalidomide (LEN) regimen demonstrated efficacy in ASCT-ineligible patients with R/R DLBCL[10]
 - After ≥35 months' follow-up, objective response rate (ORR) was 57.5% (46/80), with a complete response (CR) occurring in 40.0% of patients (32/80); median duration of response (DoR) was 43.9 months and median OS was 33.5 months[10]
- To isolate the tafasitamab contribution to the efficacy of the tafasitamab + LEN combination, a retrospective comparative effectiveness study, RE-MIND (NCT04150328)[11], generated a LEN monotherapy matched comparator cohort for L-MIND using patient-level data
- Building upon RE-MIND and the potential of real-word data (RWD) studies to enable robust efficacy comparisons with real world and clinical trial data, RE-MIND2 (NCT04697160) compared patient outcomes from L-MIND with matched patient populations treated with NCCN or European Society for Medical Oncology [ESMO]) recommended therapies for ASCT-ineligible patients with R/R DLBCL

Aim

- RE-MIND2 is a retrospective, observational cohort study designed to generate a real-world control for outcomes from the L-MIND study, to characterize the effectiveness of tafasitamab + LEN, in a real-world setting, relative to commonly administered systemic therapies for ASCT ineligible patients with R/R DLBCL

Patients and methods

- Data were collected between 1 April and 13 November 2020 using health records from academic hospitals, public hospitals, and private practices in North America, Europe, and the Asia Pacific region (Figure 1)
 - The assessment time frame for tafasitamab + LEN was March 2016 to November 2017, for systemic therapies pooled April 2010 to November 2020, May 2010 to September 2020 for BR and October 2010 to September 2020 for R-GemOx
- Eligibility criteria were based on the L-MIND study:[10] patients aged ≥18 years with histologically confirmed DLBCL and who had received ≥2 prior systemic therapies for R/R DLBCL (including ≥1 anti-CD20 therapy)
- Matching criteria and estimated propensity score (ePS)-based method were applied and efficacy outcomes from the L-MIND cohort were compared with those treated with the observational cohort of patients treated with NCCN/ESMO-listed treatment regimens enrolled in RE-MIND2 database

Figure 1. Data collection from the observational cohort

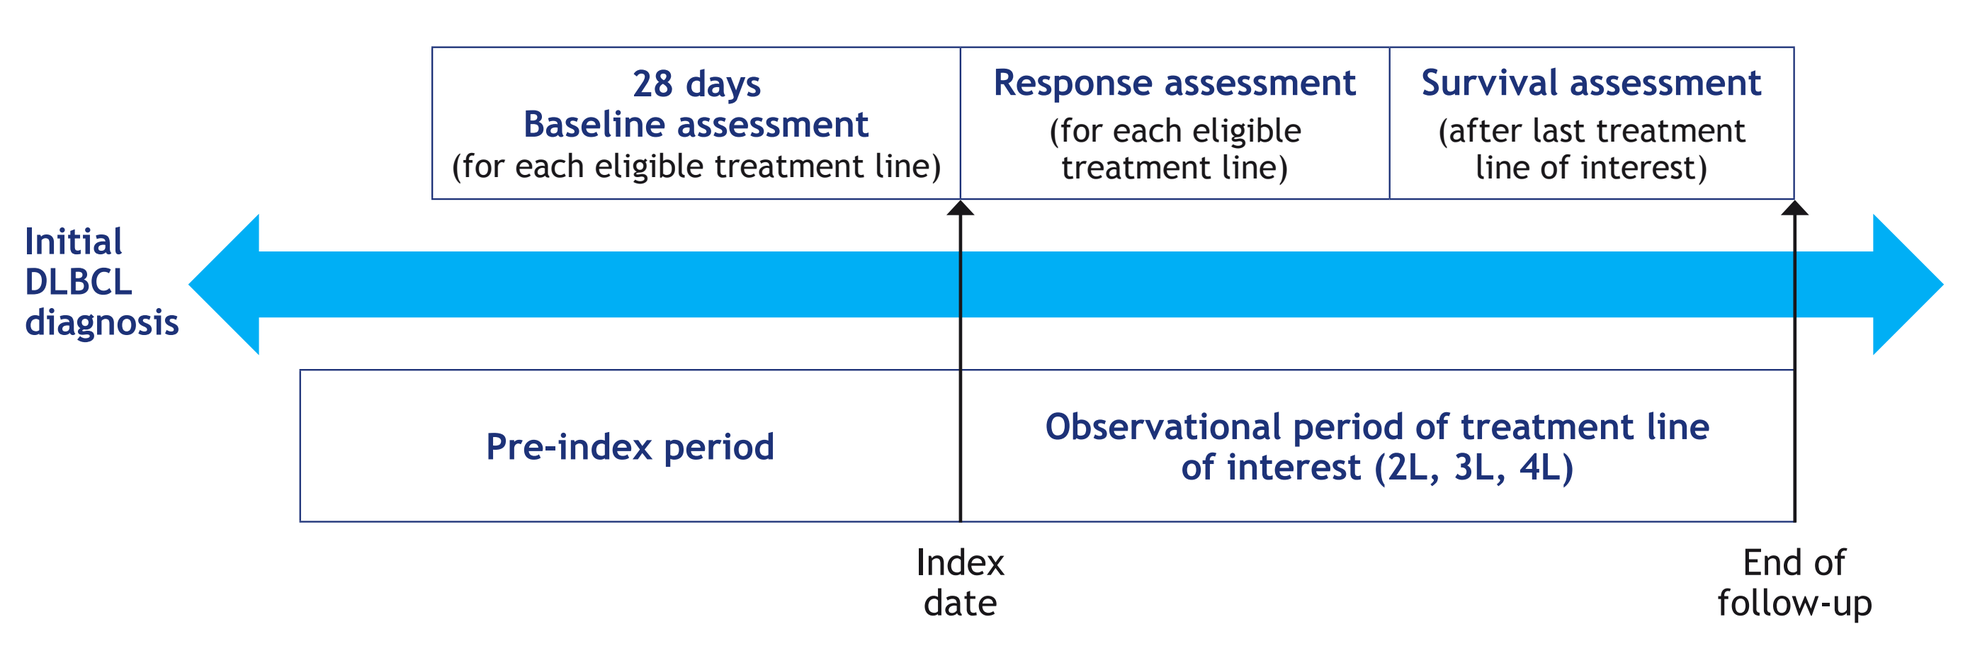
28 days
Baseline assessment
(for each eligible treatment line)
Response assessment
(for each eligible treatment line)
Survival assessment
(after last treatment line of interest)
Initial DLBCL diagnosis
Pre-index period
Observational period of treatment line of interest (2L, 3L, 4L)
Index date
End of follow-up

Patients who received at least two therapy lines for DLBCL were assigned an index date (index date 2L, 3L or 4L [i.e., second, third or fourth line]) for each eligible therapy line. Pre-index period: time between initial DLBCL diagnosis and index date of treatments (2L, 3L or 4L). Index date: start of R/R DLBCL treatment (2L, 3L or 4L). Observational period: Time between index date and end of follow-up including survival assessment. Baseline: 28 days of baseline assessment prior to index date.
DLBCL, diffuse large-B cell lymphoma; L, therapy line; R/R, relapsed/refractory.

- Three matched analysis sets (MAS) were created comprising cohorts receiving tafasitamab + LEN versus cohorts of systemic therapies pooled for R/R DLBCL, BR, and R-GemOx
- The cohorts in each MAS were matched using ePS-based 1:1 nearest neighbor (NN) matching, balanced for nine baseline prognostic covariates, as listed in Table 1. To achieve high quality of balance between cohorts, the standard mean difference of each covariate post-matching was pre-defined as ≤0.2
- The primary endpoint was OS and secondary endpoints included ORR, CR rate, progression-free survival (PFS), and DoR
- Sensitivity analysis was conducted for efficacy endpoints to establish the robustness of the comparisons
 - This analysis included additional covariates during patient matching: Eastern Cooperative Oncology Group performance status (0–1 vs ≥2), and replacing history of primary refractoriness with history of early relapse (yes vs no) and history of primary progressive (yes vs no)

Results

- In total, 3,454 patients were enrolled from 200 sites
- Following matching criteria, n=76 patients from the L-MIND study and n=961 patients in the RE-MIND2 observational cohort were selected to be eligible for matching. Patients in the observational cohort who were ASCT-eligible were excluded from matching
- The NN 1:1 matching method resulted in strictly matched pairs of patients: tafasitamab + LEN vs systemic therapies pooled (n=76 pairs), tafasitamab + LEN vs BR (n=75 pairs), and tafasitamab + LEN vs R-GemOx (n=74 pairs) (Figure 2, Table 1)
- Median duration of follow-up (months) in matched cohorts was: 31.84 vs 33.25 for tafasitamab + LEN vs systemic therapies pooled, respectively; 32.92 vs 25.00 for tafasitamab + LEN vs BR, respectively; and 32.92 vs 33.18 for tafasitamab vs R-GemOx, respectively

Figure 2. Number of patients analyzed per MAS

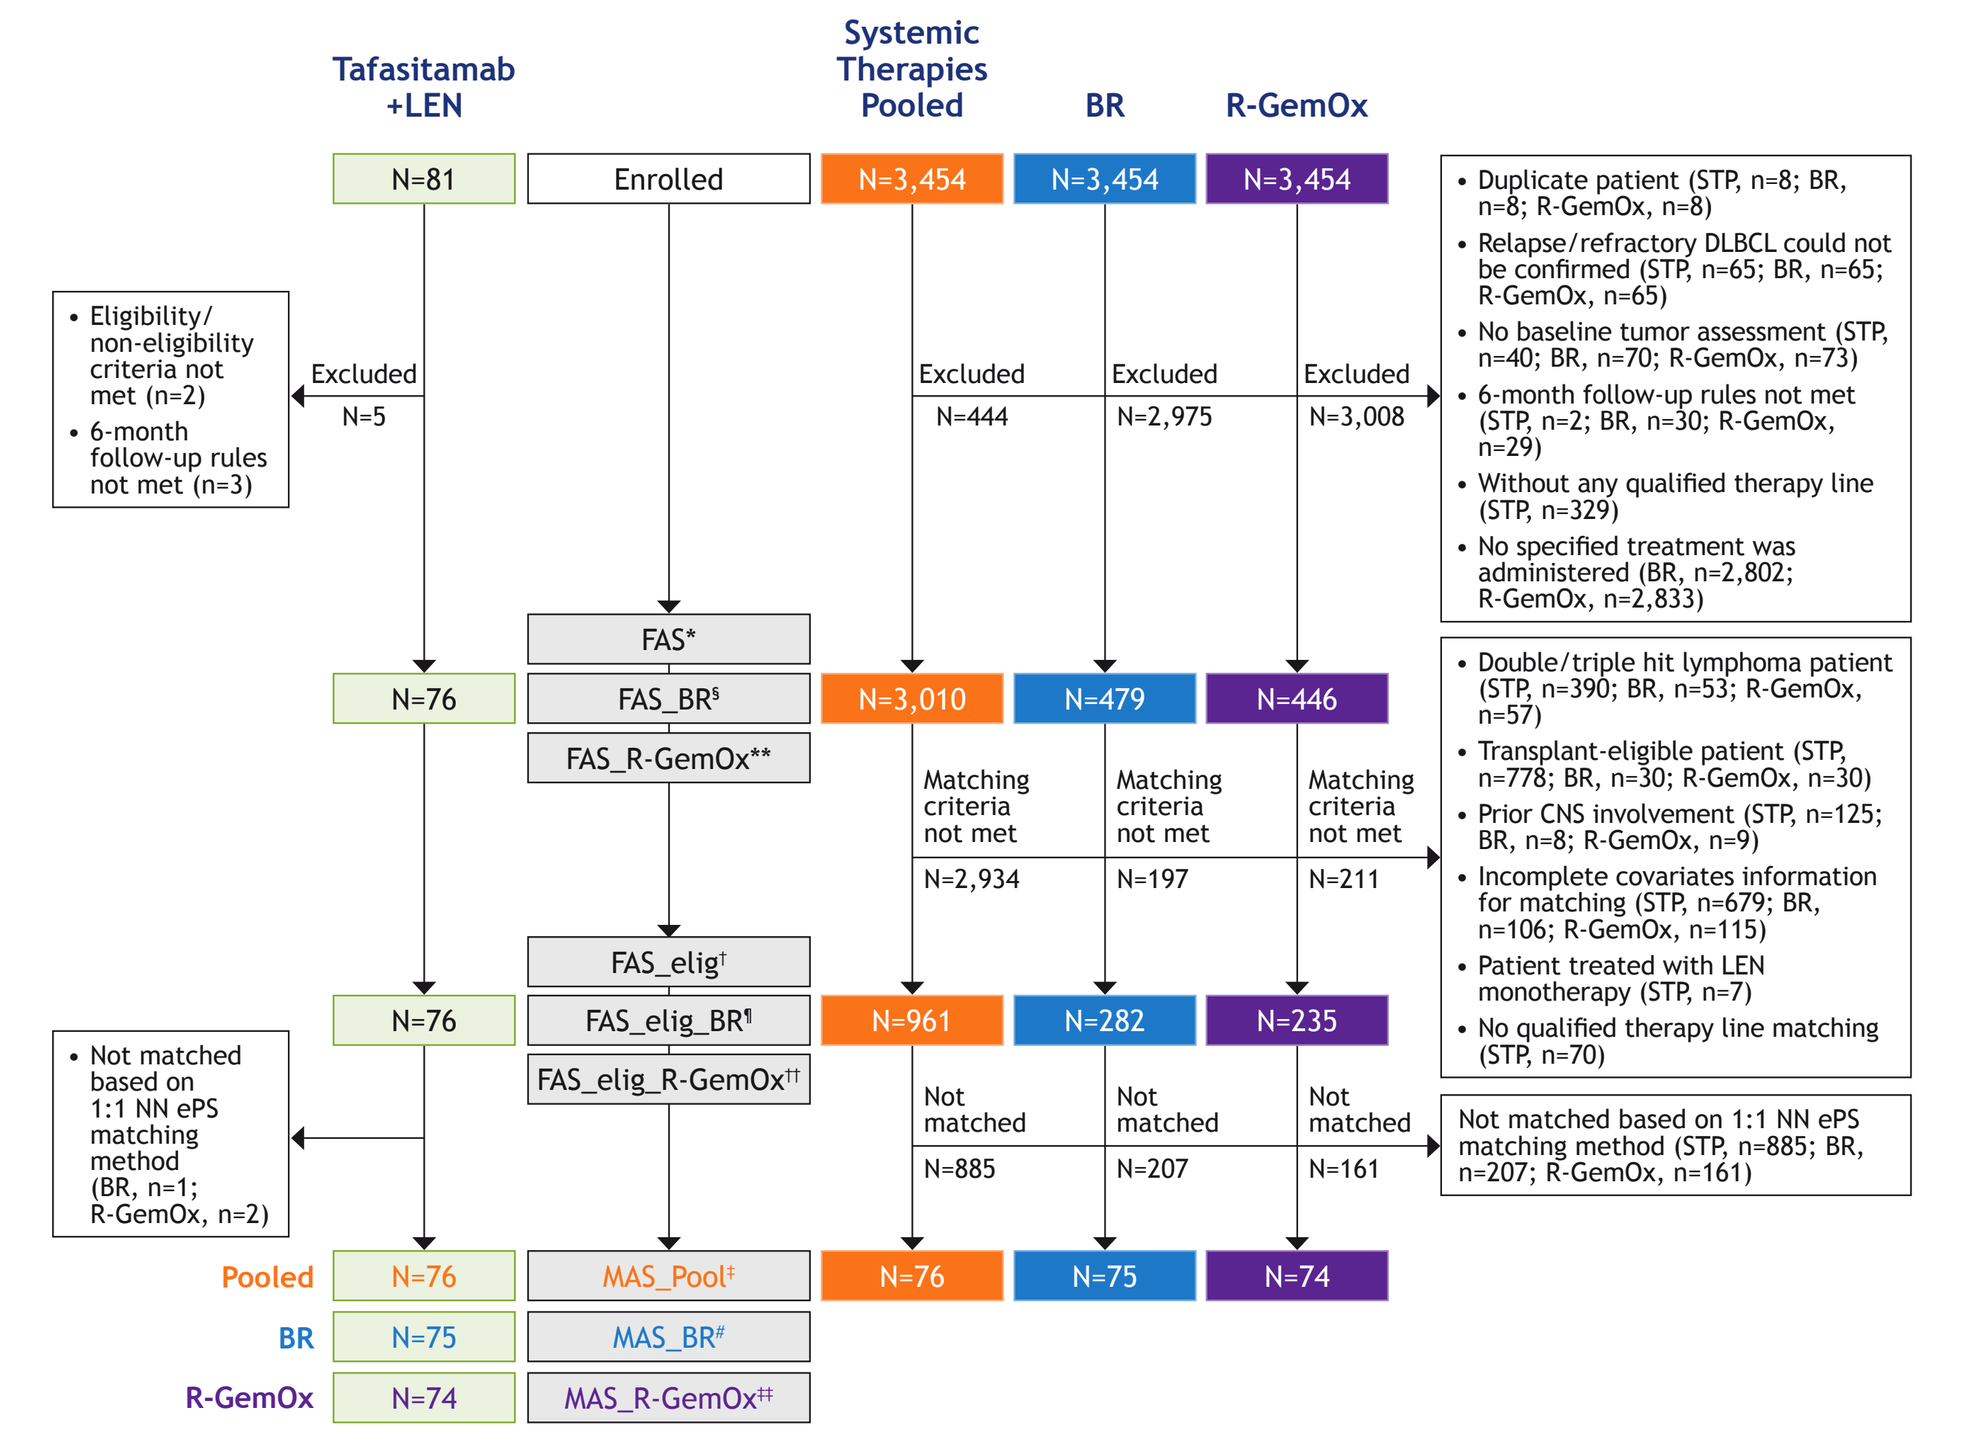
Tafasitamab +LEN
Systemic Therapies Pooled
BR
R-GemOx
N=81
Enrolled
N=3,454
N=3,454
N=3,454
• Eligibility/non-eligibility criteria not met (n=2)
• 6-month follow-up rules not met (n=3)
Excluded N=5
Excluded N=444
Excluded N=2,975
Excluded N=3,008
• Duplicate patient (STP, n=8; BR, n=8; R-GemOx, n=8)
• Relapse/refractory DLBCL could not be confirmed (STP, n=65; BR, n=65; R-GemOx, n=65)
• No baseline tumor assessment (STP, n=40; BR, n=70; R-GemOx, n=73)
• 6-month follow-up rules not met (STP, n=2; BR, n=30; R-GemOx, n=29)
• Without any qualified therapy line (STP, n=329)
• No specified treatment was administered (BR, n=2,802; R-GemOx, n=2,833)
FAS*
N=76
FAS_BR§
FAS_R-GemOx**
N=3,010
N=479
N=446
Matching criteria not met N=2,934
Matching criteria not met N=197
Matching criteria not met N=211
• Double/triple hit lymphoma patient (STP, n=390; BR, n=53; R-GemOx, n=57)
• Transplant-eligible patient (STP, n=778; BR, n=30; R-GemOx, n=30)
• Prior CNS involvement (STP, n=125; BR, n=8; R-GemOx, n=9)
• Incomplete covariates information for matching (STP, n=679; BR, n=106; R-GemOx, n=115)
• Patient treated with LEN monotherapy (STP, n=7)
• No qualified therapy line matching (STP, n=70)
FAS_elig†
N=76
FAS_elig_BR¶
FAS_elig_R-GemOx††
N=961
N=282
N=235
• Not matched based on 1:1 NN ePS matching method (BR, n=1; R-GemOx, n=2)
Not matched N=885
Not matched N=207
Not matched N=161
Not matched based on 1:1 NN ePS matching method (STP, n=885; BR, n=207; R-GemOx, n=161)
Pooled N=76 MAS_Pool‡ N=76
BR N=75 MAS_BR# N=75
R-GemOx N=74 MAS_R-GemOx‡‡ N=74

*FAS included patients who met the eligible/non-eligible criteria of RE-MIND2 and patients from the L-MIND study, all patients had a minimum of 6 months follow-up. †FAS_elig included patients who were eligible for matching. ‡MAS_Pool included 1:1 matched patients from the L-MIND study and the observational cohort using baseline covariates. §FAS_BR included patients who met the eligible/non-eligible criteria of RE-MIND2 and received BR, and patients from the L-MIND study who received at least one dose of tafasitamab and one dose of LEN; all patients had a minimum of 6 months follow-up. ¶FAS_elig_BR included a subset of patients from FAS_BR who were eligible for matching; #MAS_BR included 1:1 matched patients from the L-MIND study and those who received BR. **FAS_R-GemOx included patients who met the eligible/non-eligible criteria of RE-MIND2 and received R-GemOx, and patients from the L-MIND study who received at least one dose of tafasitamab and one dose of LEN; all patients had a minimum of 6 months follow-up. ††FAS_elig_R-GemOx included a subset of patients from FAS_R-GemOx who were eligible for matching; ‡‡MAS_R-GemOx included 1:1 matched patients from the L-MIND study and those who received R-GemOx. BR, bendamustine + rituximab; DLBCL, diffuse large-B cell lymphoma; elig, eligible; ePS, estimated propensity score; FAS, full analysis set; L, therapy line; LEN, lenalidomide; MAS, matched analysis set; R-GemOx, rituximab + gemcitabine + oxaliplatin; R/R, relapsed/refractory; STP; systemic therapies pooled.

Table 1. Demographics and baseline characteristics

Matching characteristics		MAS for systemic therapies pooled: Tafasitamab + LEN (n=76)	Systemic therapies pooled (n=76)	MAS for BR: Tafasitamab + LEN (n=75)	BR (n=75)	MAS for R-GemOx: Tafasitamab + LEN (n=74)	R-GemOx (n=74)
Age, n (%)	Age <70 years	33 (43.4)	31 (40.8)	33 (44.0)	33 (44.0)	31 (41.9)	26 (35.1)
	Age ≥70 years	43 (56.6)	45 (59.2)	42 (56.0)	42 (56.0)	43 (58.1)	48 (64.9)
Ann Arbor stage, n (%)	I+II	19 (25.0)	19 (25.0)	18 (24.0)	19 (25.3)	18 (24.3)	15 (20.3)
	III+IV	57 (75.0)	57 (75.0)	57 (76.0)	56 (74.7)	56 (75.7)	59 (79.7)
Refractoriness to last prior therapy, n (%)	Yes	34 (44.7)	35 (46.1)	33 (44.0)	32 (42.7)	33 (44.6)	29 (39.2)
	No	42 (55.3)	41 (53.9)	42 (56.0)	43 (57.3)	41 (55.4)	45 (60.8)
Number of prior systemic treatment lines, n (%)	1	39 (51.3)	39 (51.3)	39 (52.0)	39 (52.0)	39 (52.7)	41 (55.4)
	2	32 (42.1)	32 (42.1)	31 (41.3)	22 (29.3)	30 (40.5)	26 (35.1)
	3	5 (6.6)	5 (6.6)	5 (6.7)	14 (18.7)	5 (6.8)	7 (9.5)
History of primary refractoriness, n (%)	Yes	14 (18.4)	12 (15.8)	14 (18.7)	19 (25.3)	14 (18.9)	14 (18.9)
	No	62 (81.6)	64 (84.2)	61 (81.3)	56 (74.7)	60 (81.1)	60 (81.1)
Prior ASCT, n (%)	Yes	9 (11.8)	10 (13.2)	9 (12.0)	14 (18.7)	8 (10.8)	8 (10.8)
	No	67 (88.2)	66 (86.8)	66 (88.0)	61 (81.3)	66 (89.2)	66 (89.2)
Elevated LDH (>ULN), n (%)	Yes	41 (53.9)	44 (57.9)	41 (54.7)	37 (49.3)	41 (55.4)	48 (64.9)
	No	35 (46.1)	32 (42.1)	34 (45.3)	38 (50.7)	33 (44.6)	26 (35.1)
Neutropenia (cut-off <1.5 x 109/L), n (%)	Yes	2 (2.6)	2 (2.6)	2 (2.7)	4 (5.3)	2 (2.7)	5 (6.8)
	No	74 (97.4)	74 (97.4)	73 (97.3)	71 (94.7)	72 (97.3)	69 (93.2)
Anemia (cut-off hemoglobin <10 g/dL), n (%)	Yes	6 (7.9)	5 (6.6)	6 (8.0)	5 (6.7)	6 (8.1)	5 (6.8)
	No	70 (92.1)	71 (93.4)	69 (92.0)	70 (93.3)	68 (91.9)	69 (93.2)
Other characteristics							
Sex, n (%)	Female	36 (47.4)	32 (42.1)	36 (48.0)	30 (40.0)	35 (47.3)	38 (51.4)
	Male	40 (52.6)	44 (57.9)	39 (52.0)	45 (60.0)	39 (52.7)	36 (48.6)
Age at index date, years	Mean (SD)	69.1 (9.71)	68.7 (11.88)	69.0 (9.75)	69.3 (9.23)	69.5 (9.59)	71.0 (9.64)
	Median (Q1–Q3)	71.5 (62.0–76.0)	72.0 (60.0–77.0)	71.0 (62.0–76.0)	71.0 (62.0–76.0)	72.0 (62.0–76.0)	73.5 (66.0–77.0)
	Range (min–max)	41–86	37–87	41–86	48–86	41–86	40–87
ECOG PS, n (%)	0–1	70 (92.1)	44 (57.9)	69 (92.0)	40 (53.3)	68 (91.9)	37 (50.0)
	≥2	6 (7.9)	21 (27.6)	6 (8.0)	23 (30.7)	6 (8.1)	28 (37.8)
	Missing	0	11 (14.5)	0	12 (16.0)	0	9 (12.2)
Primary progressive disease, n (%)	Yes	2 (2.6)	5 (6.6)	2 (2.7)	8 (10.7)	2 (2.7)	6 (8.1)
	No	74 (97.4)	71 (93.4)	73 (97.3)	67 (89.3)	72 (97.3)	68 (91.9)

ASCT, autologous stem cell transplant; BR, bendamustine + rituximab; ECOG PS, Eastern Cooperative Oncology Group performance status; LDH, lactate dehydrogenase; LEN, lenalidomide; MAS, matched analysis set; Q1, lower quartile; Q3, upper quartile; R-GemOx, rituximab + gemcitabine + oxaliplatin; SD, standard deviation; ULN, upper limit of normal.

- Tafasitamab + LEN was associated with longer OS vs systemic therapies pooled (hazard ratio [HR]: 0.553; p=0.0076), BR (HR: 0.418; p<0.0001), and R-GemOx (HR: 0.467; p=0.0004) (Figure 3)
 - In the comparative observational cohort, second-line median OS for systemic therapies pooled, BR, and R-GemOx was 16.1, 12.0, and 16.8 months, respectively. However for tafasitamab + LEN, the median OS with second-line was not reached, indicating >50% patients were alive by end of follow-up time. Significantly improved OS for second-line therapy was observed with tafasitamab + LEN compared to systemic therapies pooled (HR: 0.502, p=0.0467), BR (HR=0.287, p=0.0002), and R-GemOx (HR=0.403, p=0.0067)
- ORR and CR rate were significantly higher for tafasitamab + LEN vs systemic therapies pooled and R-GemOx; a numerical improvement was observed for tafasitamab + LEN vs BR (Table 2)
- PFS was significantly improved in the tafasitamab + LEN cohort compared with systemic therapies pooled (12.1 vs 5.8 months; HR: 0.424; p<0.0001), BR (12.1 vs 7.9 months; HR: 0.527; p=0.0033), and R-GemOx (14.1 vs 5.1 months; HR: 0.433; p<0.0001) (Figure 4)
 - In the comparative observational cohort, second-line median PFS for systemic therapies pooled, BR, and R-GemOx was 8.0, 8.8, and 7.1 months, respectively. However for tafasitamab + LEN, the median PFS with second-line was 16.2 months. Significantly improved PFS for second-line therapy was observed with tafasitamab + LEN compared to systemic pooled (HR: 0.452; p=0.0081), BR (HR: 0.475; p=0.0155), and R-GemOx (HR: 0.466; p=0.0096)

Table 2. ORR and CR rate for tafasitamab + LEN vs systemic therapies pooled, BR, and R-GemOx

	MAS for systemic therapies pooled: Tafasitamab + LEN (n=76)	Systemic therapies pooled (n=76)	MAS for BR: Tafasitamab + LEN (n=75)	BR (n=75)	MAS for R-GemOx: Tafasitamab + LEN (n=74)	R-GemOx (n=74)
ORR, n (%) (95% CI)	51 (67.1) (55.4–77.5)	37 (48.7) (37.0–60.4)	50 (66.7) (54.8–77.1)	41 (54.7) (42.7–66.2)	51 (68.9) (57.1–79.2)	34 (45.9) (34.3–57.9)
Fisher's exact test p-value of ORR	0.032		0.181		0.007	
CR rate as best response, n (%) (95% CI)	29 (38.2) (27.2–50.0)	16 (21.1) (12.5–31.9)	29 (38.7) (27.6–50.6)	21 (28.0) (18.2–39.6)	29 (39.2) (28.0–51.2)	17 (23.0) (14.0–34.2)
Fisher's exact p-value of CR rate	0.032		0.225		0.050	

BR, bendamustine + rituximab; CI, confidence interval; CR, complete response; LEN, lenalidomide; MAS, matched analysis set; ORR, overall response rate; R-GemOx, rituximab + gemcitabine + oxaliplatin.

- DoR was significantly improved in the tafasitamab + LEN cohort compared with systemic therapies pooled (26.1 vs 6.6 months; HR: 0.323; p=0.005), BR (26.1 vs 9.2 months; HR: 0.371; p=0.0015), and R-GemOx (26.1 vs 9.5 months; HR: 0.372; p=0.0009) (Figure 5)
- Results from sensitivity analysis generally supported the results of the primary analysis sets
- Treatment discontinuation rates due to adverse events (AEs) with tafasitamab + LEN were 14.5%, 14.5%, and 15.1% in the MAS's for systemic therapies pooled, BR, and R-GemOx, respectively
 - AEs leading to treatment discontinuation for patients in the cohorts of systemic therapies pooled, BR, and R-GemOx were 6.8%, 2.8%, and 5.4%, respectively

Figure 3. KM plots of OS. (A) Tafasitamab + LEN vs systemic therapies pooled; (B) Tafasitamab + LEN vs BR; (C) Tafasitamab + LEN vs R-GemOx

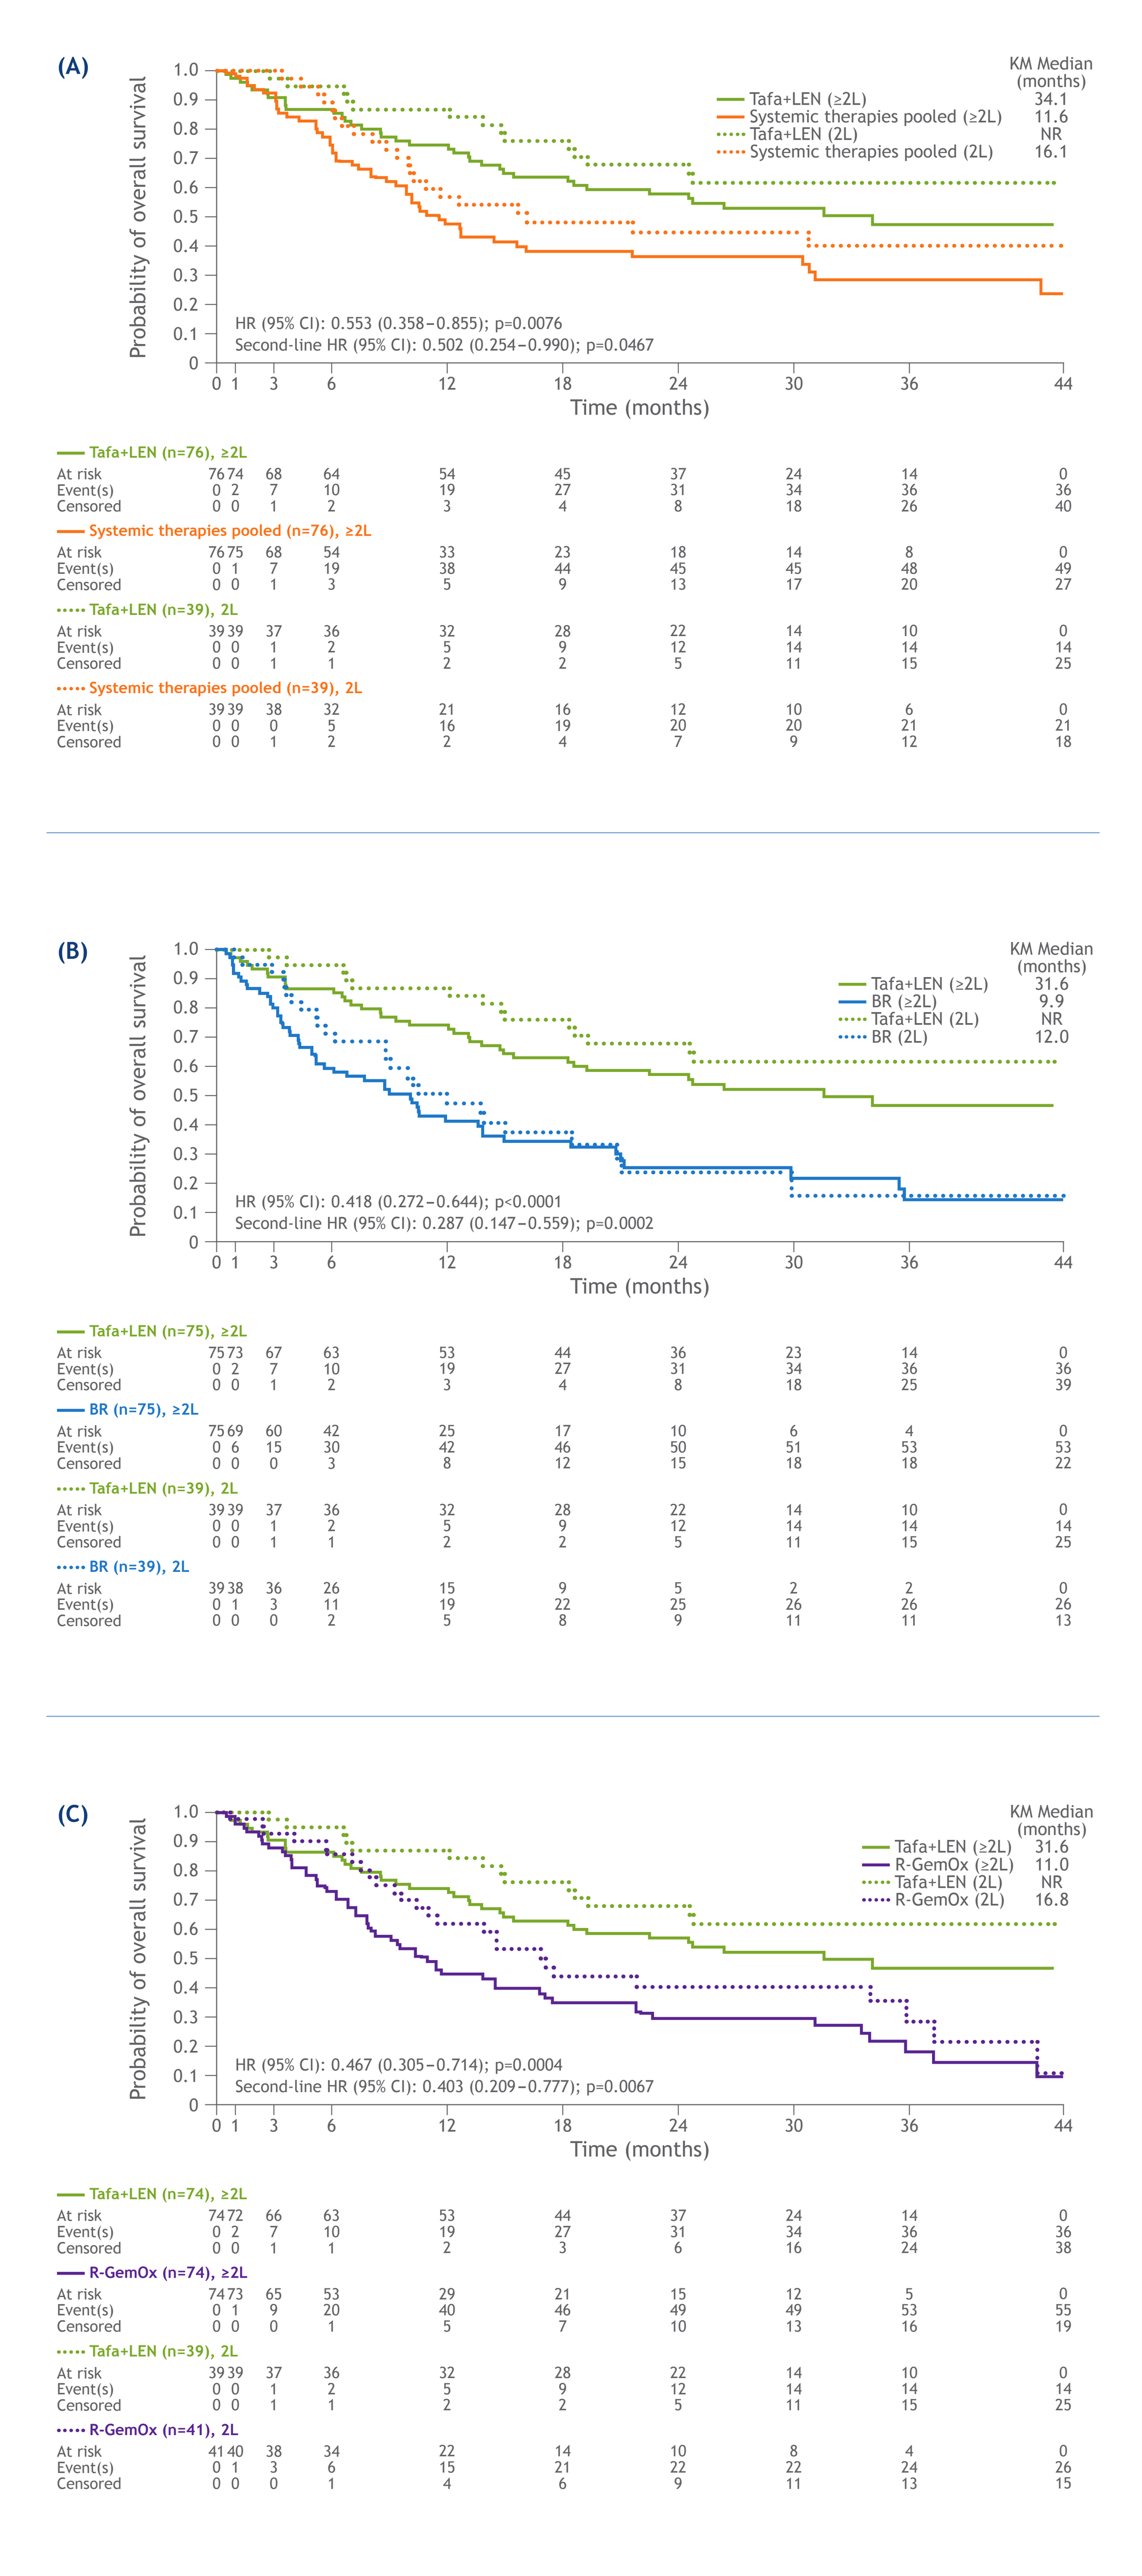
(A)
Probability of overall survival
KM Median (months)
Tafa+LEN (≥2L) 34.1
Systemic therapies pooled (≥2L) 11.6
Tafa+LEN (2L) NR
Systemic therapies pooled (2L) 16.1
HR (95% CI): 0.553 (0.358–0.855); p=0.0076
Second-line HR (95% CI): 0.502 (0.254–0.990); p=0.0467
Time (months)
(B)
KM Median (months)
Tafa+LEN (≥2L) 31.6
BR (≥2L) 9.9
Tafa+LEN (2L) NR
BR (2L) 12.0
HR (95% CI): 0.418 (0.272–0.644); p<0.0001
Second-line HR (95% CI): 0.287 (0.147–0.559); p=0.0002
(C)
KM Median (months)
Tafa+LEN (≥2L) 31.6
R-GemOx (≥2L) 11.0
Tafa+LEN (2L) NR
R-GemOx (2L) 16.8
HR (95% CI): 0.467 (0.305–0.714); p=0.0004
Second-line HR (95% CI): 0.403 (0.209–0.777); p=0.0067

BR, bendamustine + rituximab; CI, confidence interval; HR, hazard ratio; KM, Kaplan-Meier; L, therapy line; LEN, lenalidomide; NR, not reached; R-GemOx, rituximab + gemcitabine + oxaliplatin; tafa, tafasitamab.

Figure 4. KM plots of PFS. (A) Tafasitamab + LEN vs systemic therapies pooled; (B) Tafasitamab + LEN vs BR; (C) Tafasitamab + LEN vs R-GemOx

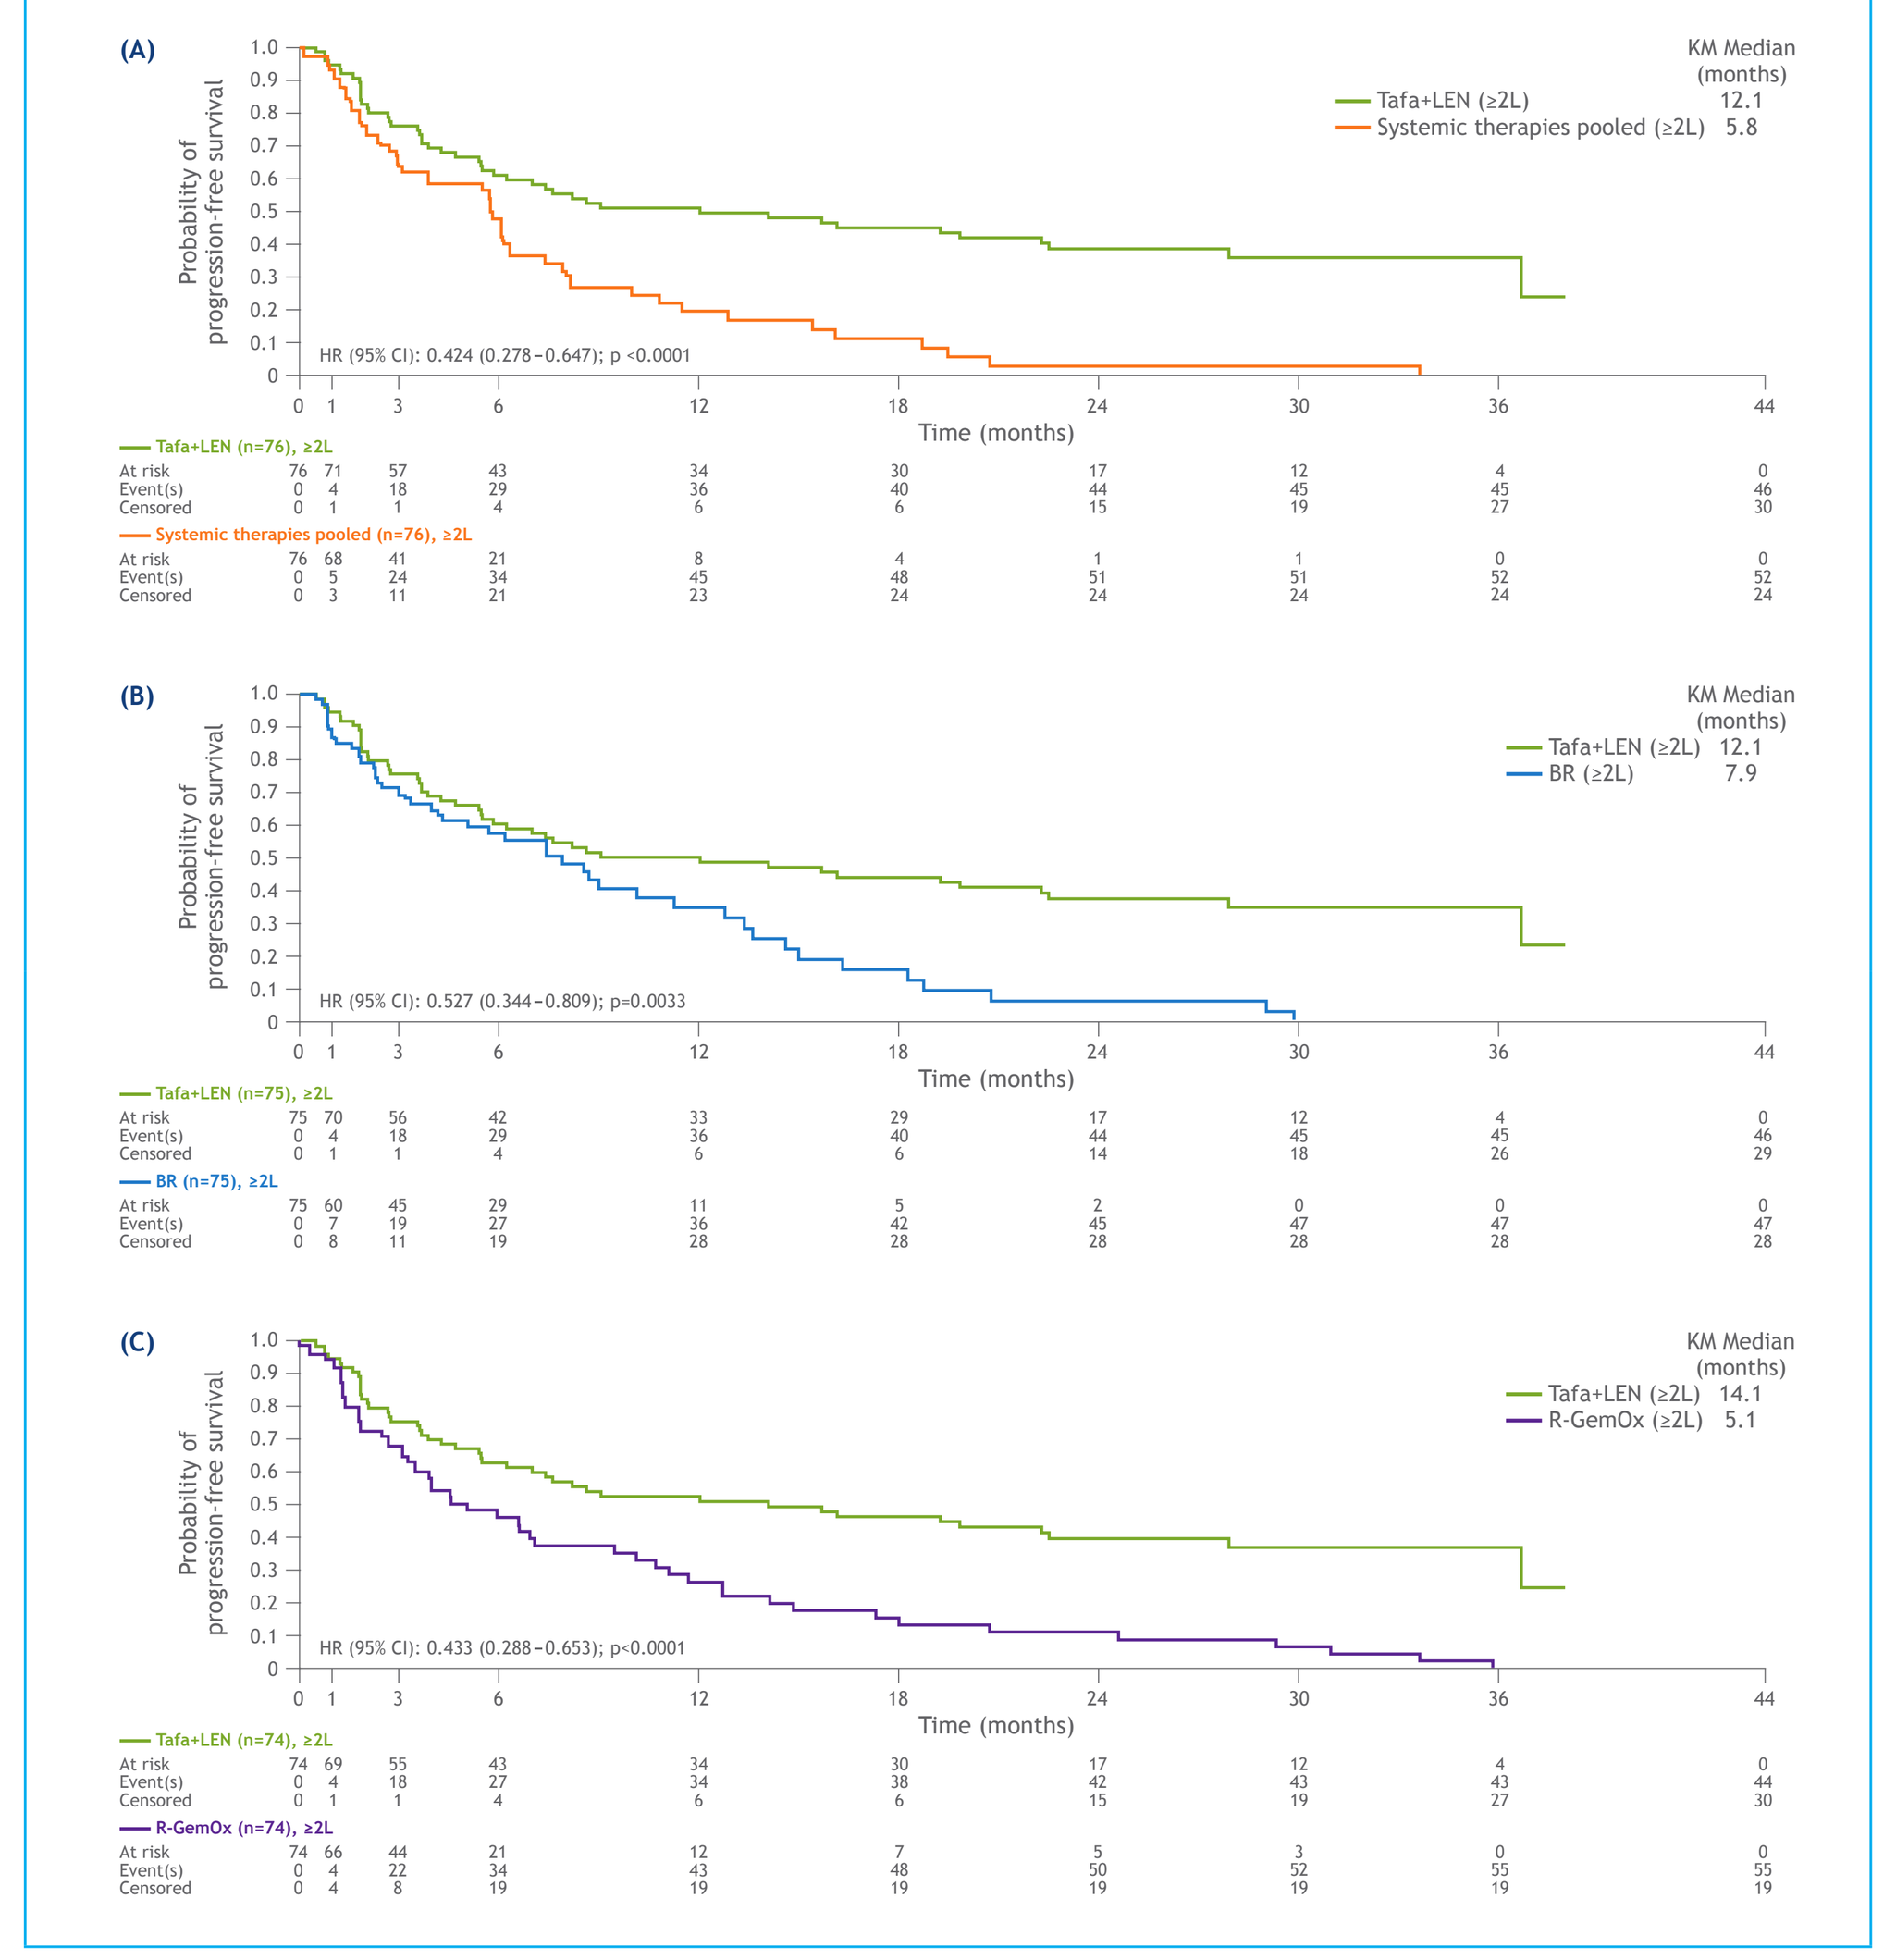
(A)
Probability of progression-free survival
KM Median (months)
Tafa+LEN (≥2L) 12.1
Systemic therapies pooled (≥2L) 5.8
HR (95% CI): 0.424 (0.278–0.647); p <0.0001
Time (months)
(B)
Tafa+LEN (≥2L) 12.1
BR (≥2L) 7.9
HR (95% CI): 0.527 (0.344–0.809); p=0.0033
(C)
Tafa+LEN (≥2L) 14.1
R-GemOx (≥2L) 5.1
HR (95% CI): 0.433 (0.288–0.653); p<0.0001

BR, bendamustine + rituximab; CI, confidence interval; HR, hazard ratio; KM, Kaplan-Meier; L, therapy line; LEN, lenalidomide; PFS, progression-free survival; R-GemOx, rituximab + gemcitabine + oxaliplatin; tafa, tafasitamab.

Figure 5. KM plots of DoR. (A) Tafasitamab + LEN vs systemic therapies pooled; (B) Tafasitamab + LEN vs BR; (C) Tafasitamab + LEN vs R-GemOx

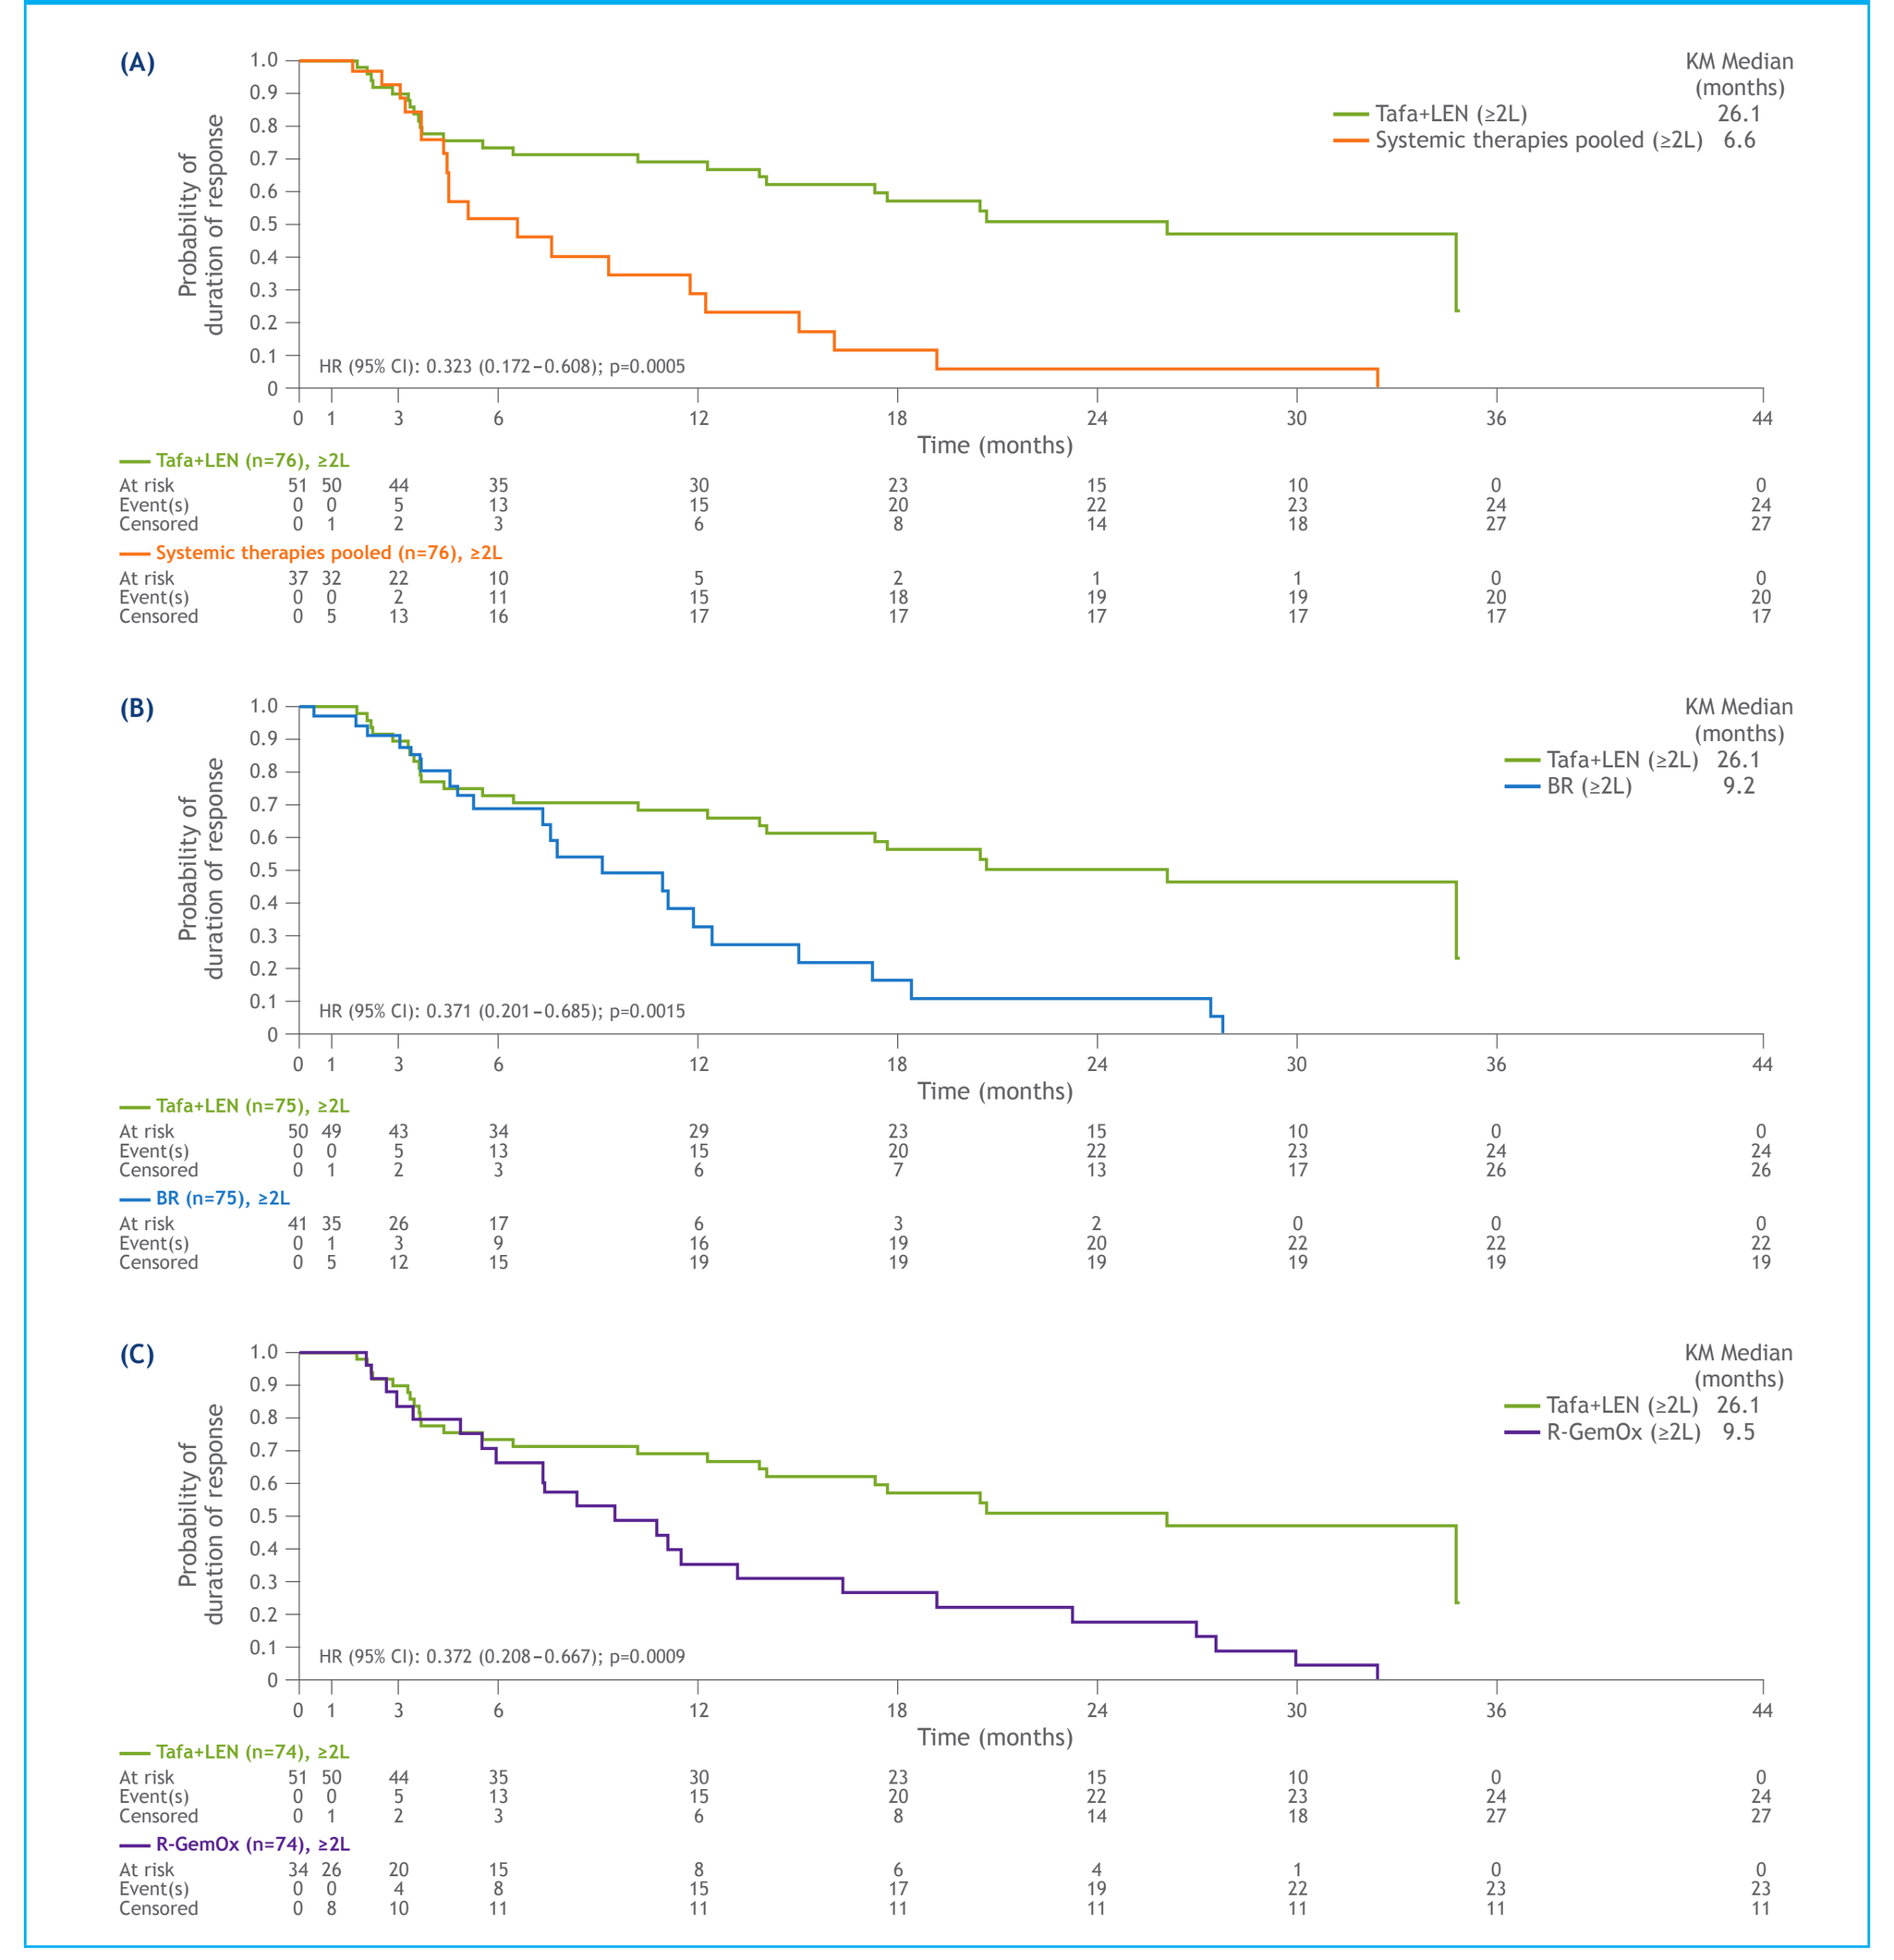
(A)
Probability of duration of response
KM Median (months)
Tafa+LEN (≥2L) 26.1
Systemic therapies pooled (≥2L) 6.6
HR (95% CI): 0.323 (0.172–0.608); p=0.0005
Time (months)
(B)
Tafa+LEN (≥2L) 26.1
BR (≥2L) 9.2
HR (95% CI): 0.371 (0.201–0.685); p=0.0015
(C)
Tafa+LEN (≥2L) 26.1
R-GemOx (≥2L) 9.5
HR (95% CI): 0.372 (0.208–0.667); p=0.0009

BR, bendamustine + rituximab; DoR, duration of response; KM, Kaplan-Meier; L, therapy line; LEN, lenalidomide; R-GemOx, rituximab + gemcitabine + oxaliplatin; tafa, tafasitamab.

Discussion

- The primary endpoint of the study was met with a statistically significant improvement in OS for the tafasitamab + LEN cohort versus matched observational cohorts of systemic therapies pooled, BR, and R-GemOx
- In addition, results from the present study align with data reported from previous studies on BR and R-GemOx (Table 3)

Table 3. Overview of BR and R-GemOx results reported in literature vs RE-MIND2 study

	Key studies with BR: Literature-reported outcomes			RE-MIND2 outcomes	Key studies with R-GemOx: Literature-reported outcomes				RE-MIND2 outcomes
Reference:	12	13	8		14	9	15	16	
N	59	59	40	75	49	196	32	46	74
ORR, %	62.7	45.8	25	54.7	61	38	78	83	45.9
CR, %	37.3	15.3	22.5	28.0	44	33	50	50	23.0
mPFS, months	6.7	3.6	3.7	7.9	5	5	NA	NA	5.1
mOS, months	NA	NA	4.7	9.9	11	10	NA	NA	11.0

BR, bendamustine + rituximab; CR, complete response; mOS, median overall survival; mPFS, median progression-free survival; NA, not available; ORR, overall response rate; R-GemOx, rituximab + gemcitabine + oxaliplatin.

- In the comparative observational cohort, second-line median OS in the tafasitamab + LEN cohort was not reached, indicating that >50% of second-line patients in this group were alive by the end of the follow-up time
- To reduce bias, and ensure robust and accurate comparisons, a sensitivity analysis was performed using multiple baseline covariates; providing additional covariate balance

Conclusions

- RE-MIND2 was designed to generate a real-world control for outcomes from the L-MIND study to characterize the effectiveness of tafasitamab + LEN relative to other systemic therapies, currently recommended for the treatment of ASCT-ineligible patients with R/R DLBCL, using a 1:1 NN ePS-based matching method
- Tafasitamab + LEN was associated with longer OS vs systemic therapies pooled, BR, and R-GemOx with an HR of 0.553, 0.418, and 0.467, respectively
- Overall, results of this study show that this immunomodulatory, chemotherapy-free regimen may improve outcomes compared with NCCN/ESMO-recommended therapies used in routine clinical care for the treatment of R/R DLBCL
- As large randomized trials in R/R DLBCL are limited, real-world data can be used to compare efficacy in well-designed studies with matching for multiple covariates

Acknowledgments

This study was funded by MorphoSys AG. Medical writing assistance was provided by Ifigeneia Mavragani, PhD, of Syneos Health, UK, and funded by MorphoSys AG.

Disclosures

About Tafasitamab

Tafasitamab is a humanized, Fc-modified, cytolytic CD19-targeting monoclonal antibody. In 2010, MorphoSys licensed exclusive worldwide rights to develop and commercialize tafasitamab from Xencor, Inc. Tafasitamab incorporates an XmAb® engineered Fc domain, which mediates B-cell lysis through apoptosis and immune effector mechanism, including antibody-dependent cell-mediated cytotoxicity (ADCC) and antibody-dependent cellular phagocytosis (ADCP). In January 2020, MorphoSys and Incyte entered into a collaboration and licensing agreement to further develop and commercialize tafasitamab globally. Following accelerated approval by the US Food and Drug Administration in July 2020, tafasitamab is being co-commercialized by MorphoSys and Incyte in the United States. Incyte has exclusive commercialization rights outside the United States. XmAb® is a trademark of Xencor, Inc.

Conflicts of interest

GSN: consulting/advisory fees from Celgene, MorphoSys, Genentech, Selvita, Debiopharm Group, Kite/Gilead; research funding from Celgene, NanoString Technologies, MorphoSys. **DHY, PM:** nothing to disclose. **EJ:** consulting/advisory fees from Astra Zeneca, Epizyme. **AP:** consulting/advisory fees from AbbVie, Incyte. **IF:** consulting/advisory fees from Abbvie, Astra Zeneca, Bristol-Myers Squibb, Celgene, Janssen, Kite-Gilead, Merck, Novartis, Roche, Seattle Genetics. **RG:** honoraria from Celgene, Roche, Merck, Takeda, Astra Zeneca, Novartis, Amgen, Bristol-Meyer Squibb, MSD, Sandoz, AbbVie, Gilead, Daiichi Sankyo; meeting/travel support from Roche, Amgen, Janssen, Astra Zeneca, Novartis, MSD, Celgene, Gilead, Bristol-Meyer Squibb, AbbVie, Daiichi Sankyo; research funding from Celgene, Roche, Merck, Takeda, Astra Zeneca, Novartis, Amgen, Bristol-Meyers Squibb, MSD, Sandoz, Abbvie, Gilead, Daiichi Sankyo; consulting/advisory fees from Celgene, Novartis, Roche, Bristol-Myers Squibb, Takeda, Abbvie, Astra Zeneca, Janssen, MSD, Merck, Gilead, Daiichi Sankyo. **MK:** consulting/advisory fees from Antegene, Genor BioPharma, Roche. **RM:** consulting/advisory fees from Merck, Kite/Gilead; honoraria from Kite/Gilead; meeting/travel support from AbbVie. **KK:** research funding from Astra Zeneca. **PLZ:** consulting/advisory fees from Verastem, Celltrion, Gilead, Janssen-Cilag, Bristol-Meyer Squibb, Servier, Sandoz, MSD, TG Therapeutics, Takeda, Roche, EUSA Pharma, Kyowa Kirin, Novartis, ADC Therapeutics; speakers' bureau at Verastem, Celltrion, Gilead, Janssen-Cilag, Bristol-Meyer Squibb, Servier, MSD, TG Therapeutics, Takeda, Roche, EUSA Pharma, Kyowa Kirin, Novartis. **JT:** research funding from Beigene, Celgene, Bristol-Myers Squibb, Roche, Janssen, Pharmacyclics. **DH, EEW:** employees of MorphoSys. **MW:** employee of MorphoSys with stock or stock options. SA: employee of MorphoSys; travel support from MorphoSys. **GH:** research funding from Roche, Celgene, Janssen, Incyte, AbbVie, Gilead/Kite; speaker honoraria/advisory fees from Janssen, Genmab, Gilead, Astra Zeneca, Roche, Incyte, AbbVie, EUSA Pharma. **GS:** consulting/advisory fees from AbbVie, BeiGene, Bristol-Myers Squibb/Celgene, Debiopharm, Epizyme, Genentech/Roche, Genmab, Incyte, Janssen, Kite/Gilead, Miltenyi, MorphoSys, Novartis, Regeneron, VelosBio.

References

1. World Health Organization. IARC Press 2020; **2.** NCCN Guidelines®. 2021;v4; **3.** Maliske SM, et al. Blood 2019;134:2896; **4.** Sehn LH, et al. N Engl J Med 2021;384(9):842-58; **5.** González-Barca E, et al. Bone Marrow Transplant 2020;553:93–9; **6.** Chihara D, et al. Biol Blood Marrow Transplant 2014;20(5):684–9; **7.** Herrera AF, et al. J Clin Oncol 2017;3524–3; **8.** Sehn LH, et al. J Clin Oncol 2019;38(2):155–65; **9.** Cazelles C, et al. Leuk Lymphoma 2021;25; **10.** Duell J, et al. Haematologica 2021;Epub ahead of print. https://doi.org/10.3324/haematol.2020.275958; **11.** Nowakowski G, et al. J Clin Oncol 2020;38(15 Suppl):8020; **12.** Ohmachi K, et al. J Clin Oncol 2013;312103–9; **13.** Vacirca JL, et al. Ann Hematol 2014;93(3):403–9; **14.** Mounier N, et al. Haematologica 2013;98(11):1726–31; **15.** Corazzelli G, et al. Cancer Chemother Pharmacol 2009;64(5):907–16; **16.** El Gnaoui T, et al. Ann Oncol 2007;18(8):1363–8.

Presented at the Society of Hematologic Oncology (SOHO) 2021 Annual Meeting, September 8–11, 2021.

Correspondence: Prof. MD Grzegorz S. Nowakowski (Nowakowski.Grzegorz@mayo.edu)