SUBOPTIMAL VISUALIZATION IN MINIMALLY INVASIVE SURGERY - ASSESSING IMPACT THROUGH VETERAN AFFAIRS' HEALTHCARE FAILURE MODES AND EFFECTS ANALYSIS METHODOLOGY

The University of Texas at Austin
Dell Medical School

Noah Beinart[1]; Sriya Dommaraju[2]; Juslyn Dhingra[2]; John M Uecker, MD[3,4]; Christopher R Idelson, PhD[3];

[1]The McCombs School of Business, The University of Texas at Austin; [2]The College of Natural Sciences, The University of Texas at Austin; [3]ClearCam Inc., [4]Dell Medical School, Department of Surgery and Perioperative Care

Introduction

- **Clinical Background:** Clear visualization is essential for safety and efficiency in Minimally Invasive Surgery (MIS).
- **The Problem:** Image quality is constantly compromised by lens fogging, smoke, and debris.
- **Behavior Gap:** Surgeons often fight through suboptimal visual conditions to maintain workflow.
- **Consequence:** Suboptimal visualization occurs in 31-52% of MIS procedure time and is linked to nearly 1 in 5 surgical injuries.
- **AI Imperative:** Emerging AI tools require clear video to operate. Visual degradation dilutes AI performance, rendering high-tech tools potentially unsafe.

Objective

To proactively quantify the risks associated with impaired visualziation in MIS using the Veteran Affairs' Healthcare Failure Mode and Effects Analysis (HFMEA).

Methods and Materials

- **Framework:** Applied the VA National Center for Patient Safety HFMEA methodology to the intraoperative workflow.
- **Team:** A multidisciplinary team (surgeon, engineer, and research trainees) mapped the process from scope insertion to closure.
- **Risk Scoring:**
 - Identified Failure Modes for every process step.
 - Scored severity (S) and Probability (P) to calculate Hazard Score (S * P = HAZ).
- **Threshold:** Failure Modes with HAZ ≥ 8 were high risk, triggering mandatory action planning.

Results

- **The Bottleneck:** The risk analysis focused on the recurring loop of Step 4: Managing Impaired Visualization.
- **High Average Risk:** The average Hazard Score for all subprocesses of step 4 was 7.5.
- **Critical Failure Modes:** Two notably high-risks modes with a Hazard Score of 12 were identified where operations are performed under suboptimal visualization.
- **Safety Link:** These high risk events are most critical because they are directly linked to patient injury.

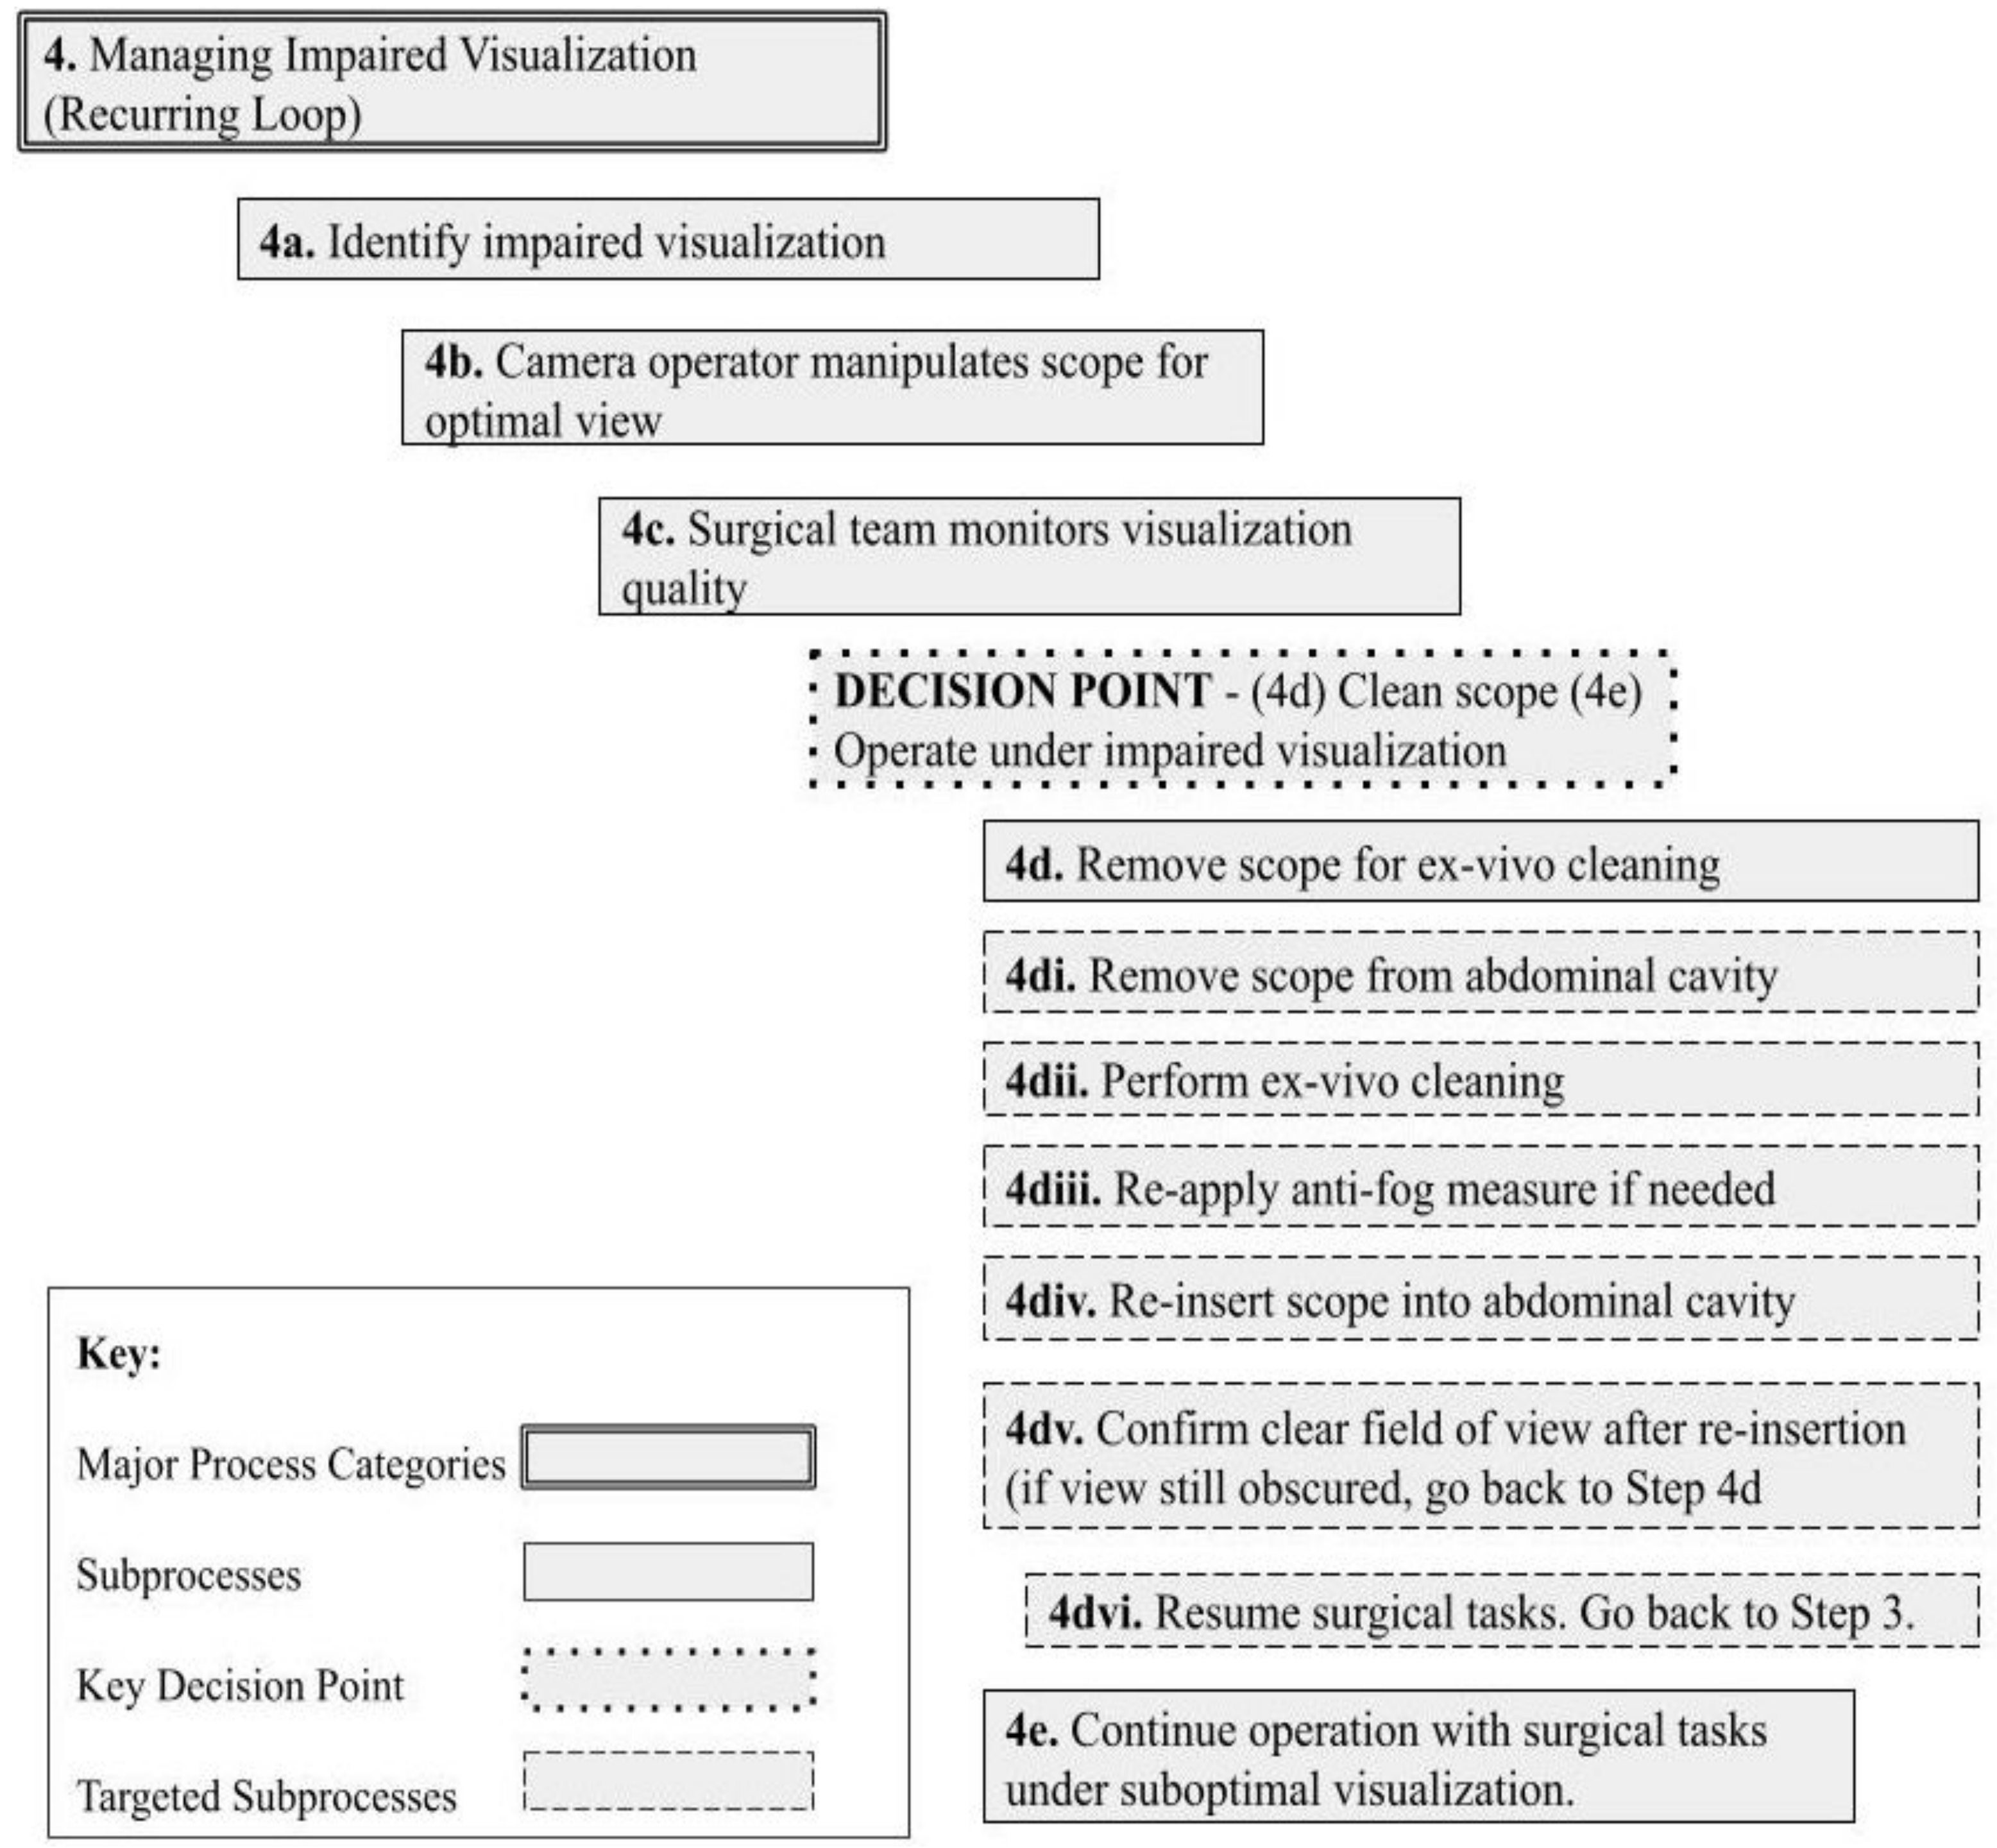
4. Managing Impaired Visualization (Recurring Loop)
4a. Identify impaired visualization
4b. Camera operator manipulates scope for optimal view
4c. Surgical team monitors visualization quality
DECISION POINT - (4d) Clean scope (4e) Operate under impaired visualization
4d. Remove scope for ex-vivo cleaning
4di. Remove scope from abdominal cavity
4dii. Perform ex-vivo cleaning
4diii. Re-apply anti-fog measure if needed
4div. Re-insert scope into abdominal cavity
4dv. Confirm clear field of view after re-insertion (if view still obscured, go back to Step 4d
4dvi. Resume surgical tasks. Go back to Step 3.
4e. Continue operation with surgical tasks under suboptimal visualization.
Key:
Major Process Categories
Subprocesses
Key Decision Point
Targeted Subprocesses

Figure 1: MIS Visualization Process Map of Step 4. The analysis focused on the critical, recurring loop in Phase 4: Managing Impaired Visualization. This cycle creates a major procedural bottleneck.

Process Step	# Failure Modes	Hazard Score (HAZ)				
		Cumulative HAZ	Avg	StDev	Max	Min
4a	3	24	8	1.7	9	6
4b	4	28	7	2.4	9	4
4c	3	30	10	1.7	12	9
4di	3	16	5.3	1.2	6	4
4dii	2	8	4	0.0	4	4
4diii	2	10	5	1.4	6	4
4div	1	8	8	N/A	8	8
4dv	1	8	8	N/A	8	8
4dvi	1	8	8	N/A	8	8
4e	1	12	12	N/A	12	12
TOTAL	21	152	7.5	-	12	4

Table 1: HFMEA Hazard Scoring Matrix. Failure modes involving "Performance under suboptimal visualization (4e) yielded the highest hazard score of 12, driven by the high probability of occurrence and sever potential for patient injury.

Discussion

- **Interpreting the Data:** The concentration of high HAZ scores within "Managing Impaired Visualization" identifies this workflow as a systemic failure point.
- **Hidden Risk:** The highest hazard scores of 12 occurred when surgeons began to tolerate critical impairment to avoid the cleaning loop to maintain surgical flow.
- **HFMEA Action Planning:** To mitigate these high risk steps, the interdisciplinary team discussed interventions of standardized trainings for visual thresholds and technological interventions..

Conclusion

- **Reframing Safety:** This proactive risk assessment suggests that lens clarity should be prioritized as an important variable for patient safety.
- **Workflow Optimization:** Reducing the HAZ scores requires improving the cleaning loop. Future efforts could focus on improved training or alternate cleaning methods.
- **Future Readiness:** As surgery begins implementing AI to aid procedures, a clear operative field is crucial for the safe use of AI-assisted platforms that rely on clear visualization.

Acknowledgements

- We would like to thank the VA National Center for Patient Safety for the creation of the HFMEA risk assessment framework.