RESECTABLE PANCREATIC CANCER: A POPULATION-BASED ANALYSIS OF PROGNOSTIC FACTORS AND OUTCOMES IN 38,796 PATIENTS

Authors: Abdul Waheed, MD; Yesenia Gonzalez, MD; Noor Nama, FCPS; Richard Hodges, MD; **Subhasis Misra, MD**

Introduction

• Pancreatic Cancer is the **4th leading cause of cancer death** in the United States

• Prognosis remains **poor despite surgical advances**

• **Survival predictors in resectable pancreatic cancer (RPC)** remain unclear

Objective

To Identify **demographic, tumor, and treatment factors** influencing survival

BayCare | St. Joseph's Hospital
General Surgery Residency Program

Methods

• **Data source**:
•Surveillance, Epidemiology, and End Results Program (SEER) database

• Study period: **1976–2016**

• **38,796 patients** with resectable pancreatic cancer identified

• **Variables analyzed**
– Age
– Sex
– Race
– Tumor location & size
– Tumor grade & histology
– Lymph node status
– Treatment modalities

• **Statistical analysis**
– Chi-square test
– Independent t-test
– Multivariate Cox regression
– Actuarial survival analysis

• Software used: **IBM SPSS Statistics**

Results

• **38,796** resectable pancreatic cancer patients identified
• **Tumor location**
– **Head:** 62.4%
– Tail: 14.7%
– Body: 7.6%
• **Pathology**
– **Moderately differentiated (Grade 2): 37%**
– **Adenocarcinoma NOS:** 47.2%
• Treatment
– Surgery performed in all patients
– **Chemotherapy:** 49.3%
– **Radiation:** 28%
• Lymph nodes
– **>4 nodes retrieved:** 55.7%
– **Node positive:** 57.9%
• Survival
– **5-year survival:** 21%
– Worst survival with **adenosquamous carcinoma (10.8%)**

Conclusion

• PC occurs most commonly in the **pancreatic head** and in **Caucasian males**
•**Surgical resection remains the mainstay of treatment**
•**Adjuvant therapy showed no clear survival benefit**