Efficacy and safety of fezolinetant in moderate-to-severe vasomotor symptoms associated with menopause

PP.0164

Johnson K,[1] Nappi RE,[2] Neal-Perry G,[3] Shapiro M C.M.,[4] Stute P,[5] Thurston RC,[6] Wolfman W,[4] English M,[7] Franklin C,[7] Lee M,[7] Santoro N[8]

[1]iResearch Atlanta LLC, Decatur, GA, USA; [2]University of Pavia, Fondazione Policlinico IRCCS S. Matteo, Pavia, Italy; [3]UNC School of Medicine, Chapel Hill, NC, USA; [4]University of Toronto, Toronto, ON, Canada; [5]Inselspital, Bern, Switzerland; [6]University of Pittsburgh School of Medicine, Pittsburgh, PA, USA; [7]Astellas Pharma Global Development, Northbrook, IL, USA; [8]University of Colorado School of Medicine, Aurora, CO, USA

INTRODUCTION

- Vasomotor symptoms (VMS), characterised by hot flashes and/or night sweats, affect many women during menopausal transition[1]
- Severe VMS adversely affect sleep, concentration, mood, energy and sexual activity[2]
- Fezolinetant, a nonhormonal selective neurokinin 3 receptor antagonist that moderates neuronal activity in the hypothalamic thermoregulatory centre, is in development for the treatment of VMS associated with menopause
- Objective: to evaluate efficacy and safety of fezolinetant versus placebo on frequency and severity of moderate-to-severe VMS associated with menopause

METHODS

Study design for SKYLIGHT 2 (NCT04003142)

- Double-blind, placebo-controlled, multicentre Phase 3 study of women aged 40–65 years with moderate-to-severe VMS associated with menopause (minimum average 7 moderate-to-severe hot flashes/day; **Figure 1**)
- Those completing 12 weeks of treatment continued their fezolinetant dose; those on placebo were re-randomised to fezolinetant 30 mg or 45 mg/day

Endpoints

- Co-primary efficacy endpoints: mean change from baseline to weeks 4 and 12 in frequency and severity of moderate-to-severe VMS
- Secondary endpoints: mean change in frequency and severity of moderate and severe VMS each week from baseline to week 12; mean change in Patient-Reported Outcomes Measurement Information System Sleep Disturbance–Short Form 8b[3] (PROMIS SD SF 8b) Total Score from baseline to week 12
- Treatment-emergent adverse events (TEAEs) assessed safety and tolerability

Statistical analysis

- For each co-primary efficacy endpoint, a mixed model repeated measures analysis of covariance model was used with change from baseline as the dependent variable and treatment group, week and smoking status (current vs former/never) as factors, with baseline measurement and baseline weight as covariates, as well as an interaction of treatment by week and an interaction of baseline measurement by week
- Statistical significance for the 4 co-primary efficacy endpoints was determined by the Hochberg method

RESULTS

Study population

- 500 women were randomised and took ≥1 dose: placebo, n=167; fezolinetant 30 mg, n=166; fezolinetant 45 mg, n=167
- Details of baseline characteristics are summarised in **Table 1**

Efficacy assessments

- Both fezolinetant doses met statistical significance in reducing VMS frequency and severity at weeks 4 and 12 versus placebo (**Figures 2 and 3**)
 - Improvements were seen as early as week 1 and maintained over 12 weeks (**Figure 4**)
- Reduction in PROMIS SD SF 8b Total Score versus placebo at week 12 was statistically significant for fezolinetant 45 mg, but not 30 mg (**Figure 5**)

Safety assessments

- A summary of the safety data is shown in **Table 2**
- Headache was the most common TEAE in the fezolinetant groups
- Of 500 women, 6 had alanine aminotransferase (ALT) values >3 times upper limit of normal (ULN): 2 fezolinetant 30 mg, 3 fezolinetant 45 mg, 1 placebo. One woman on fezolinetant 30 mg had an ALT result >5 times ULN. Aspartate aminotransferase values >3 times ULN occurred in 1 fezolinetant 30 mg and 1 placebo participant
- No elevations were associated with evidence of liver functional impairment (increased bilirubin or international normalised ratio) or liver-associated symptoms

Figure 1. Study design

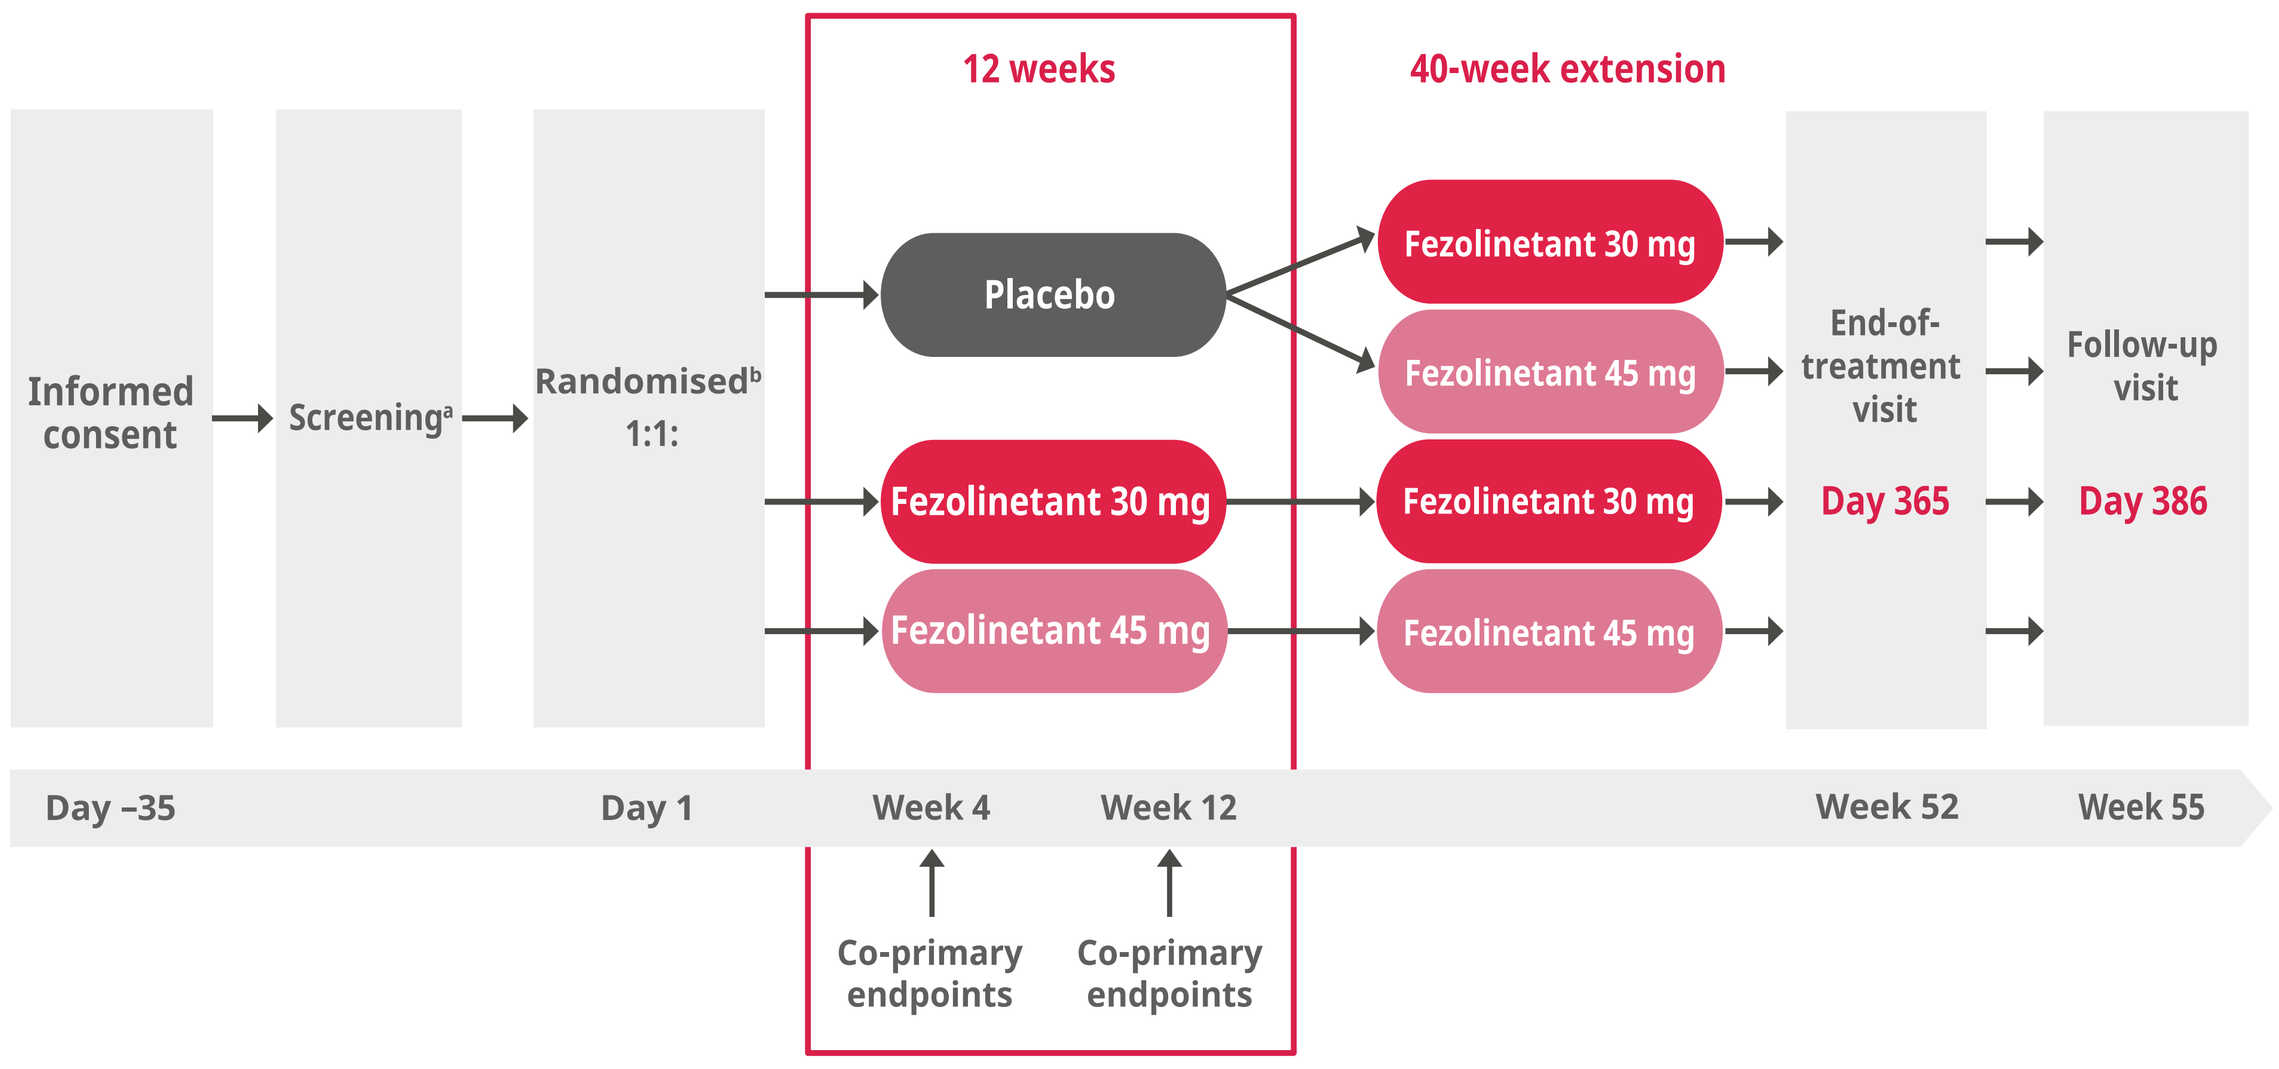
12 weeks
40-week extension
Informed consent
Screening[a]
Randomised[b] 1:1:
Placebo
Fezolinetant 30 mg
Fezolinetant 45 mg
Fezolinetant 30 mg
Fezolinetant 45 mg
Fezolinetant 30 mg
Fezolinetant 45 mg
End-of-treatment visit
Day 365
Follow-up visit
Day 386
Day –35
Day 1
Week 4
Week 12
Week 52
Week 55
Co-primary endpoints
Co-primary endpoints

FDA, US Food and Drug Administration; VMS, vasomotor symptoms

[a]VMS data were collected using an electronic hot flash diary. Women were provided with a reference guide within the diary, which included definitions for mild, moderate and severe VMS, according to FDA guidance: mild, sensation of heat without sweating; moderate, sensation of heat with sweating, able to continue activity; severe, sensation of heat with sweating, causing cessation of activity

[b]Smoking status was a randomisation stratification factor

Table 1. Demographic and baseline characteristics

	Placebo (n=167)	Fezolinetant 30 mg (n=166)	Fezolinetant 45 mg (n=167)	Total (N=500)
White, n (%)	134 (80.2)	131 (78.9)	132 (79.0)	397 (79.4)
Black/African American, n (%)	31 (18.6)	35 (21.1)	33 (19.8)	99 (19.8)
American Indian/Alaska Native, Asian, Other[a], n (%)	2 (1.2)	0	2 (1.2)	4 (0.8)
Hispanic or Latina, n (%)	32 (19.3)[b]	34 (20.5)	41 (24.6)	107 (21.4)[b]
Age, mean (SD), y	54.7 (4.6)	53.9 (4.9)	54.3 (5.4)	54.3 (5.0)
BMI, mean (SD), kg/m^2	28.16 (4.99)	27.94 (4.69)	27.91 (4.35)	28.00 (4.68)
Time since onset of HF, mean (SD), mo	81.9 (73.3)	76.2 (74.4)	81.7 (74.1)	80.0 (73.9)
Hysterectomy, n (%)	51 (30.5)	53 (31.9)	56 (33.5)	160 (32.0)

BMI, body mass index; HF, hot flash; SD, standard deviation

[a]More than 1 race

[b]Missing=1

Table 2. Overview of adverse events

TEAE, n (%)	Placebo (n=167)	Fezolinetant 30 mg (n=166)	Fezolinetant 45 mg (n=167)
TEAE	54 (32.3)	67 (40.4)	60 (35.9)
Drug-related TEAE	11 (6.6)	24 (14.5)	25 (15.0)
Serious TEAE[a]	0	3 (1.8)	2 (1.2)
Drug-related serious TEAE	0	0	0
TEAE leading to permanent discontinuation of study drug	1 (0.6)	2 (1.2)	5 (3.0)[b]
Drug-related TEAE leading to permanent discontinuation of study drug	0	1 (0.6)	5 (3.0)
TEAE by preferred term (≥2% for any group)			
Upper respiratory tract infection	7 (4.2)	5 (3.0)	5 (3.0)
Headache	4 (2.4)	5 (3.0)	6 (3.6)
Dry mouth	0	4 (2.4)	4 (2.4)
Arthralgia	1 (0.6)	5 (3.0)	1 (0.6)
Diarrhoea	4 (2.4)	1 (0.6)	2 (1.2)
Nasopharyngitis	4 (2.4)	3 (1.8)	0
Nausea	0	3 (1.8)	4 (2.4)
Weight increased	1 (0.6)	5 (3.0)	1 (0.6)

TEAE, treatment-emergent adverse event

[a]No serious TEAEs of liver enzyme elevation were reported

[b]One participant had alanine aminotransferase increased

Figure 2. Change from baseline in mean frequency of VMS

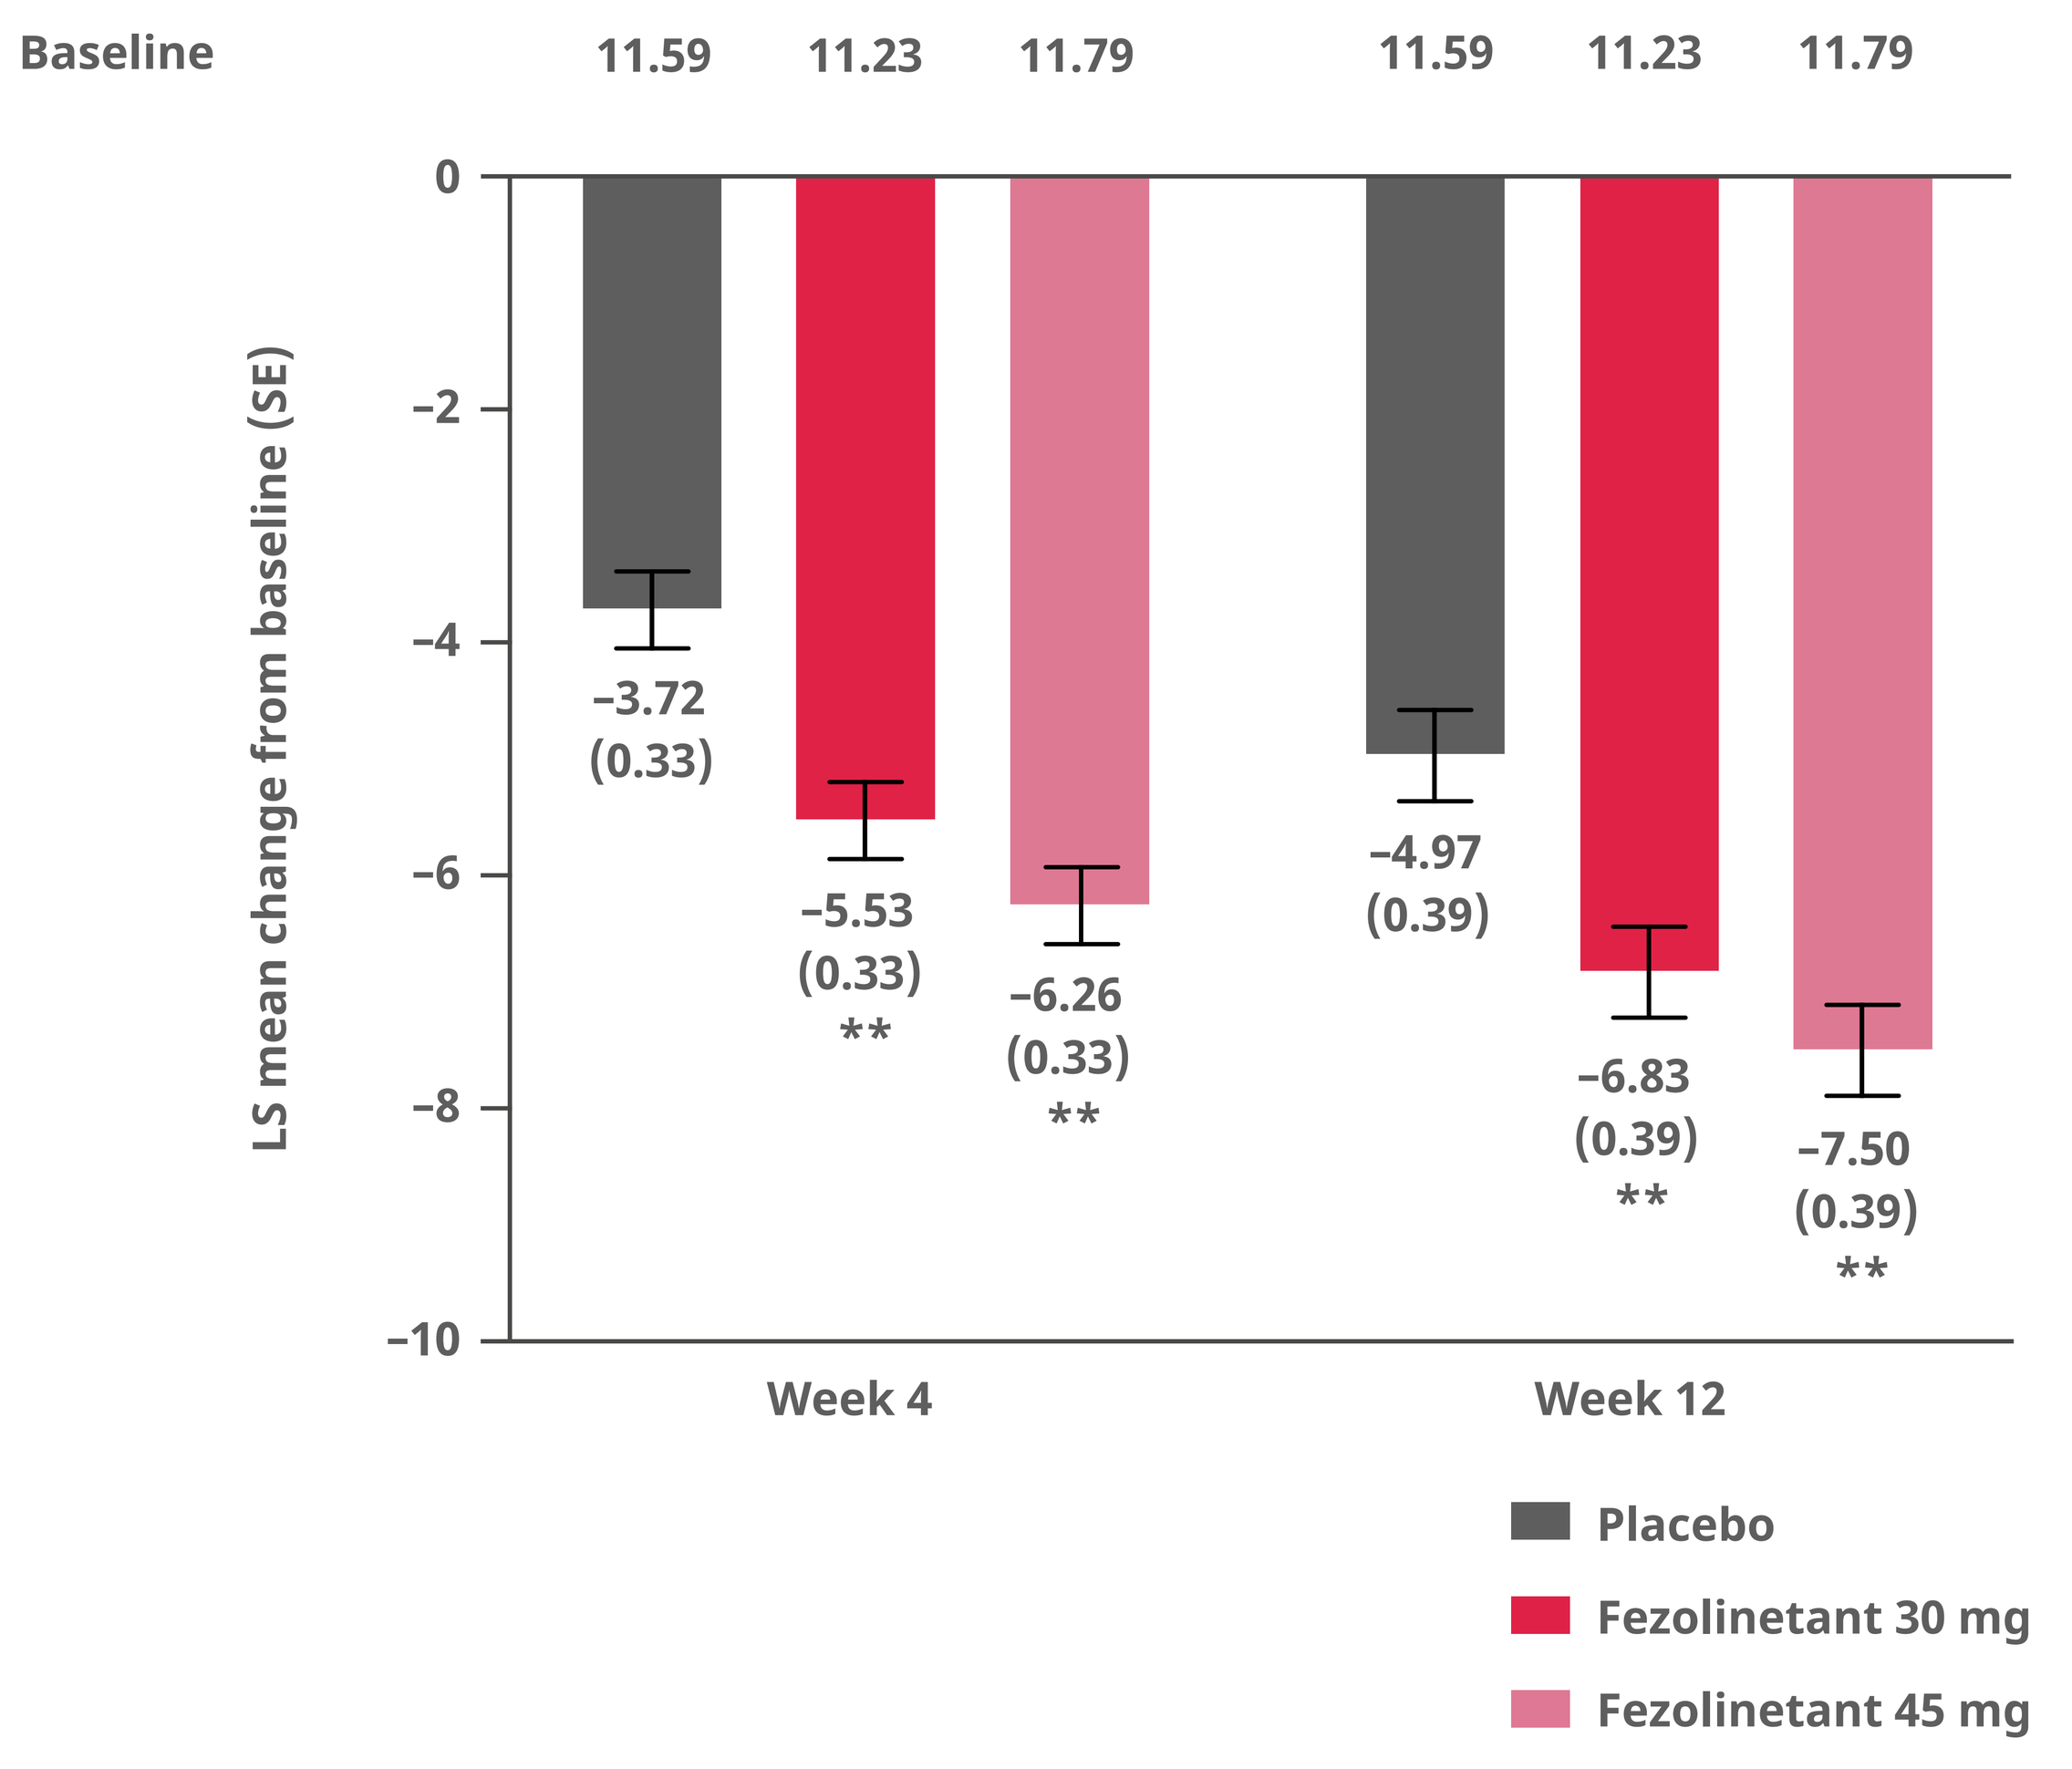
Baseline
11.59 11.23 11.79 11.59 11.23 11.79
LS mean change from baseline (SE)
0
–2
–4
–6
–8
–10
–3.72 (0.33)
–5.53 (0.33) **
–6.26 (0.33) **
–4.97 (0.39)
–6.83 (0.39) **
–7.50 (0.39) **
Week 4
Week 12
Placebo
Fezolinetant 30 mg
Fezolinetant 45 mg

LS, least squares; SE, standard error; VMS, vasomotor symptoms

**Nominal p<0.001. Statistically significant with the Hochberg method

Figure 3. Change from baseline in mean severity of VMS

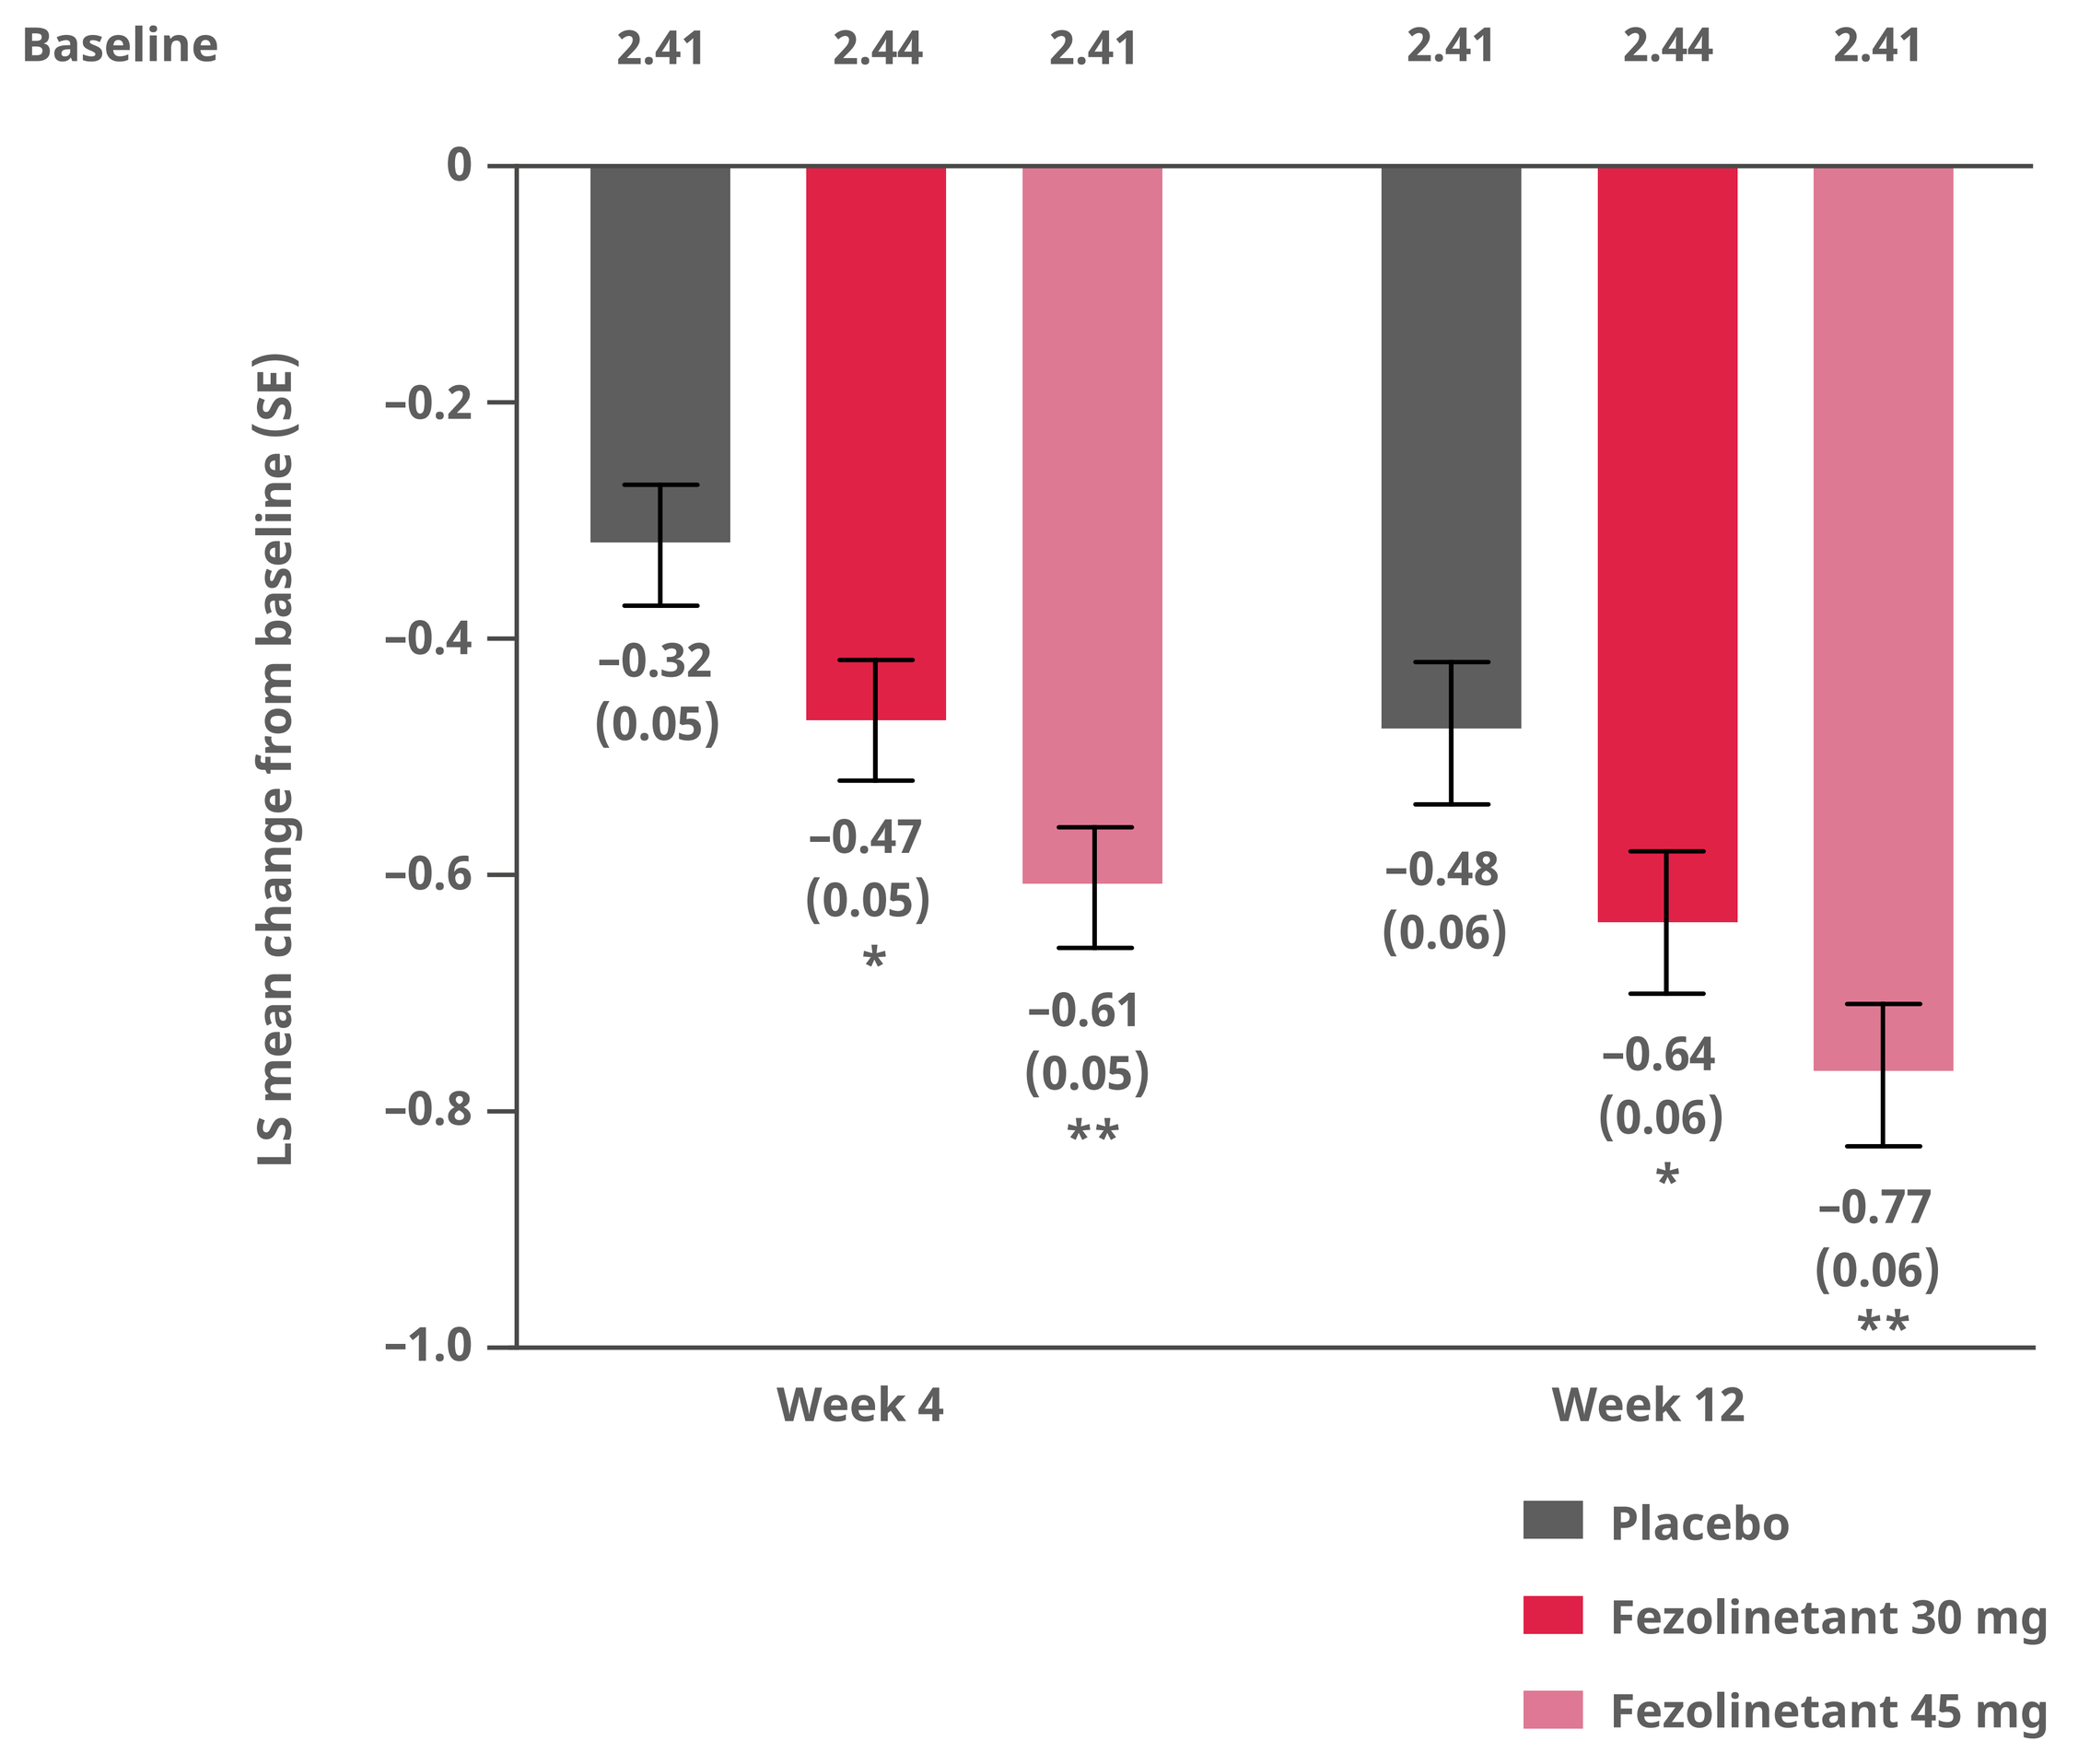
Baseline
2.41 2.44 2.41 2.41 2.44 2.41
LS mean change from baseline (SE)
0
–0.2
–0.4
–0.6
–0.8
–1.0
–0.32 (0.05)
–0.47 (0.05) *
–0.61 (0.05) **
–0.48 (0.06)
–0.64 (0.06) *
–0.77 (0.06) **
Week 4
Week 12
Placebo
Fezolinetant 30 mg
Fezolinetant 45 mg

LS, least squares; SE, standard error; VMS, vasomotor symptoms

*Nominal p<0.05; **p<0.001. Statistically significant with the Hochberg method

Figure 4. Mean frequency and severity of moderate and severe VMS over time

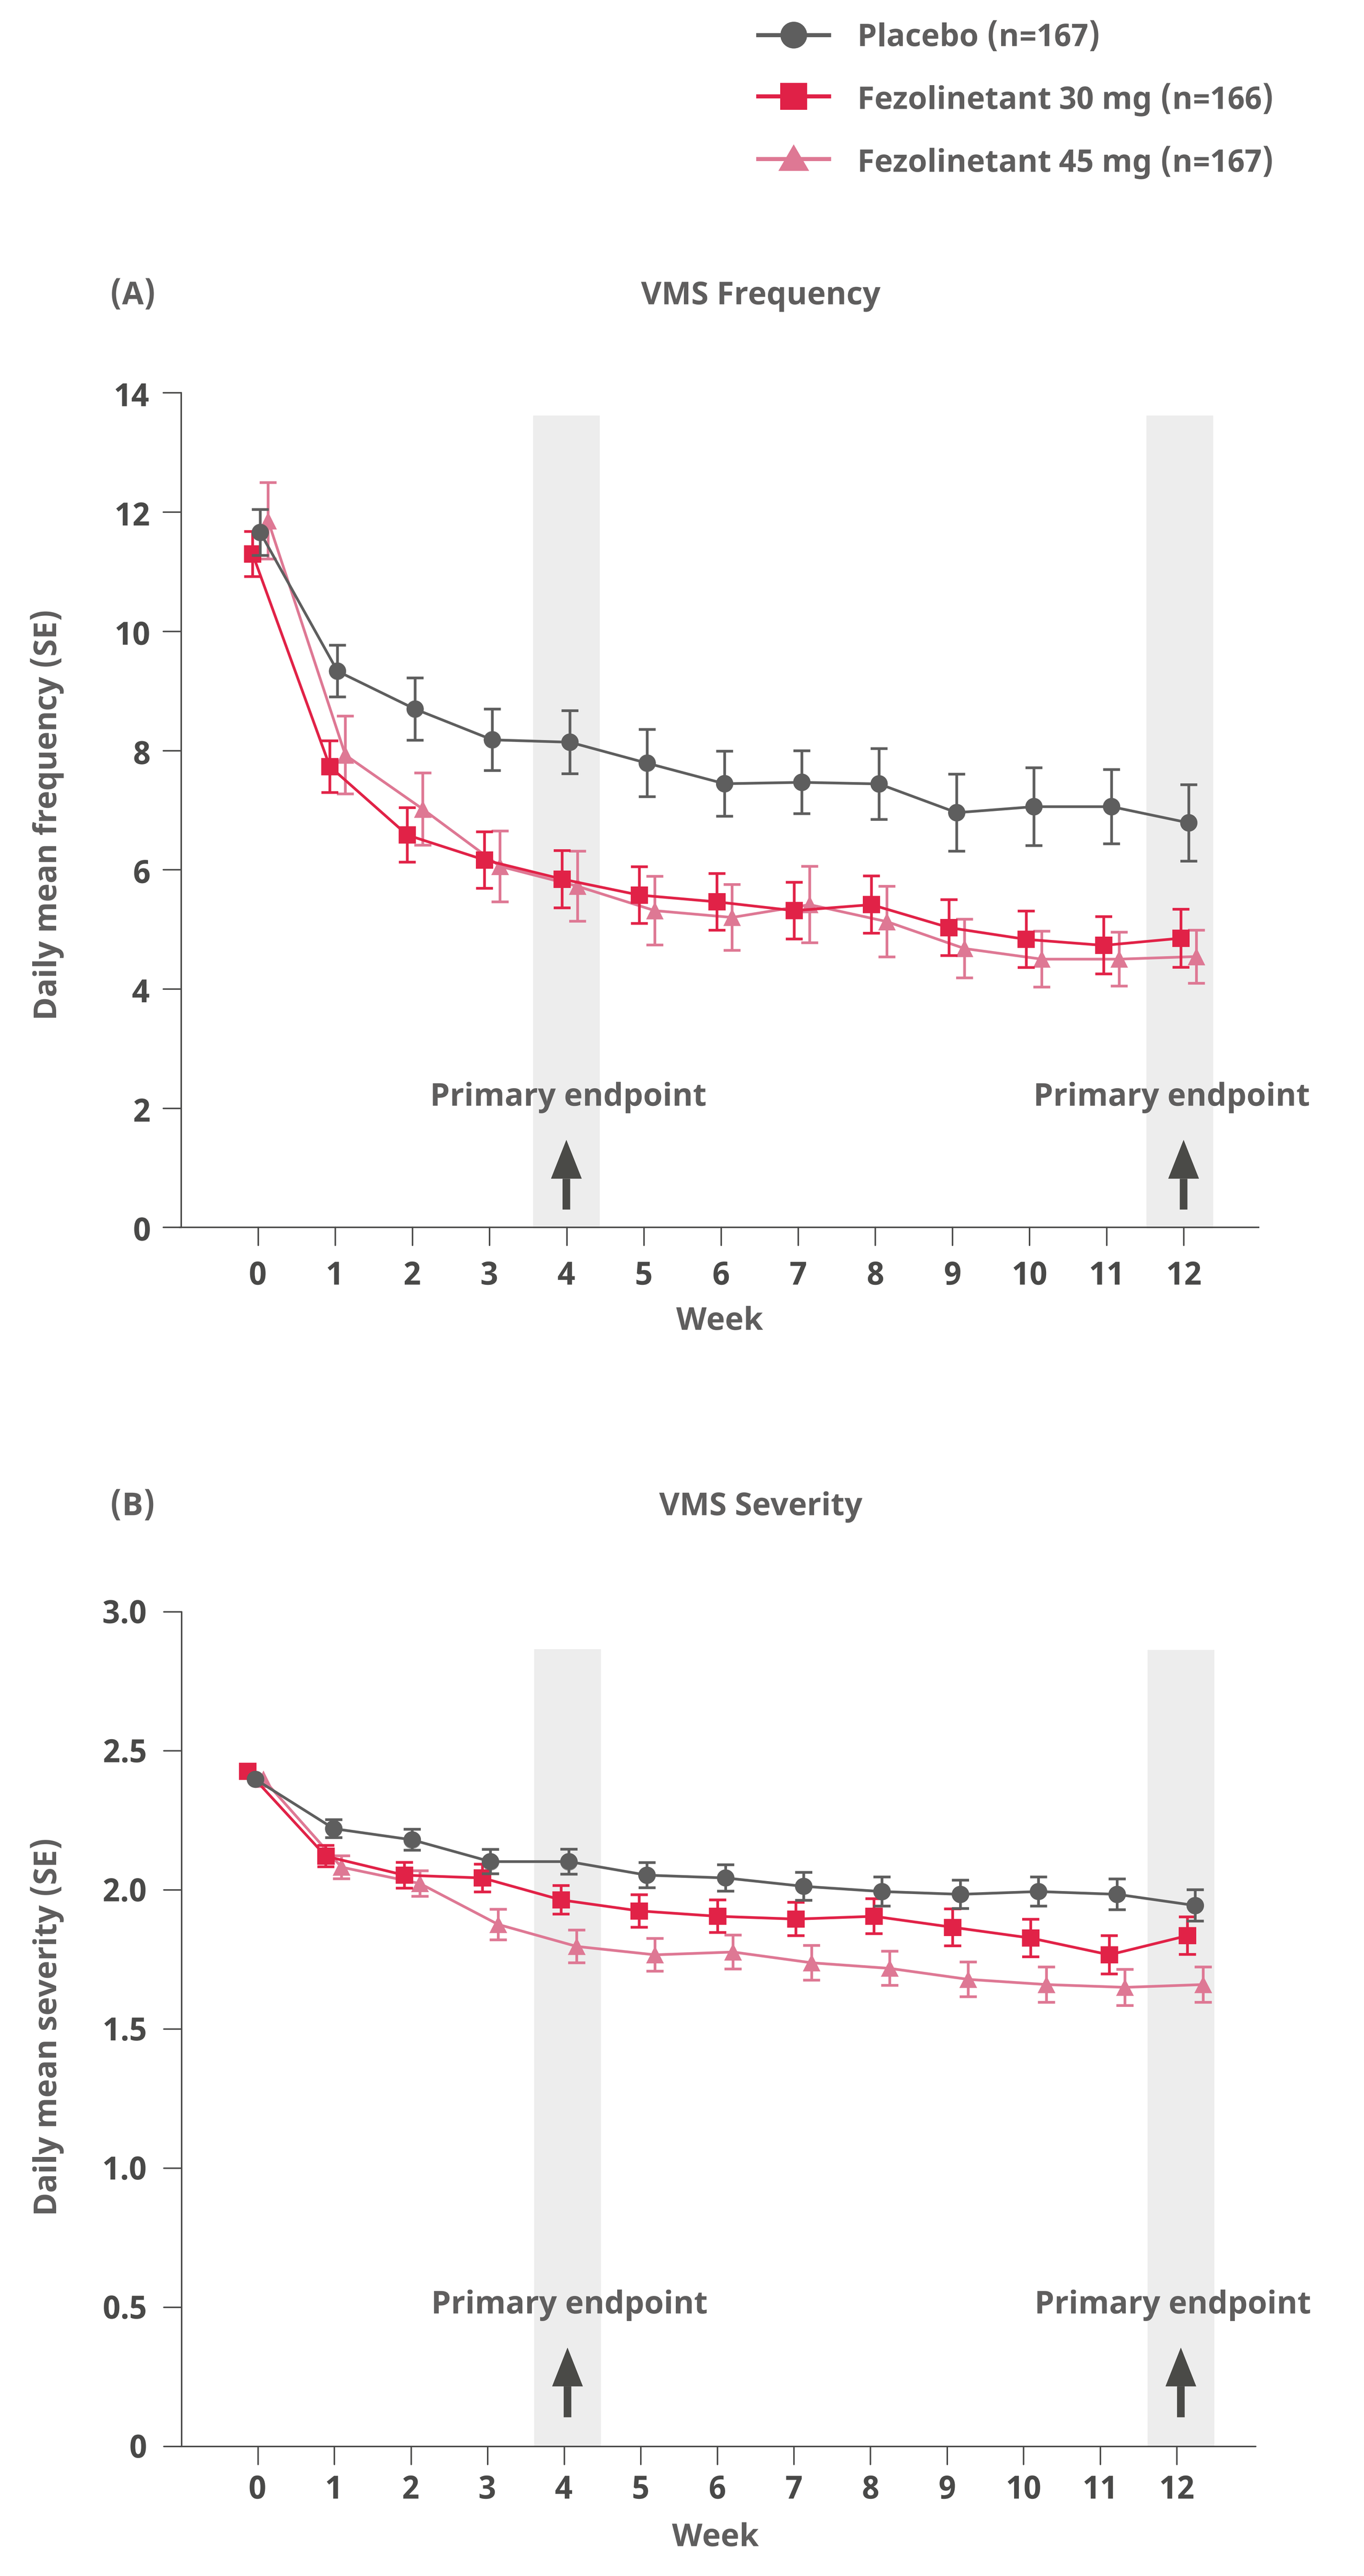
Placebo (n=167)
Fezolinetant 30 mg (n=166)
Fezolinetant 45 mg (n=167)
(A)
VMS Frequency
Daily mean frequency (SE)
14
12
10
8
6
4
2
0
Primary endpoint
Primary endpoint
0 1 2 3 4 5 6 7 8 9 10 11 12
Week
(B)
VMS Severity
Daily mean severity (SE)
3.0
2.5
2.0
1.5
1.0
0.5
0
Primary endpoint
Primary endpoint
0 1 2 3 4 5 6 7 8 9 10 11 12
Week

SE, standard error; VMS, vasomotor symptoms

Figure 5. Change from baseline in PROMIS SD SF 8b Total Score

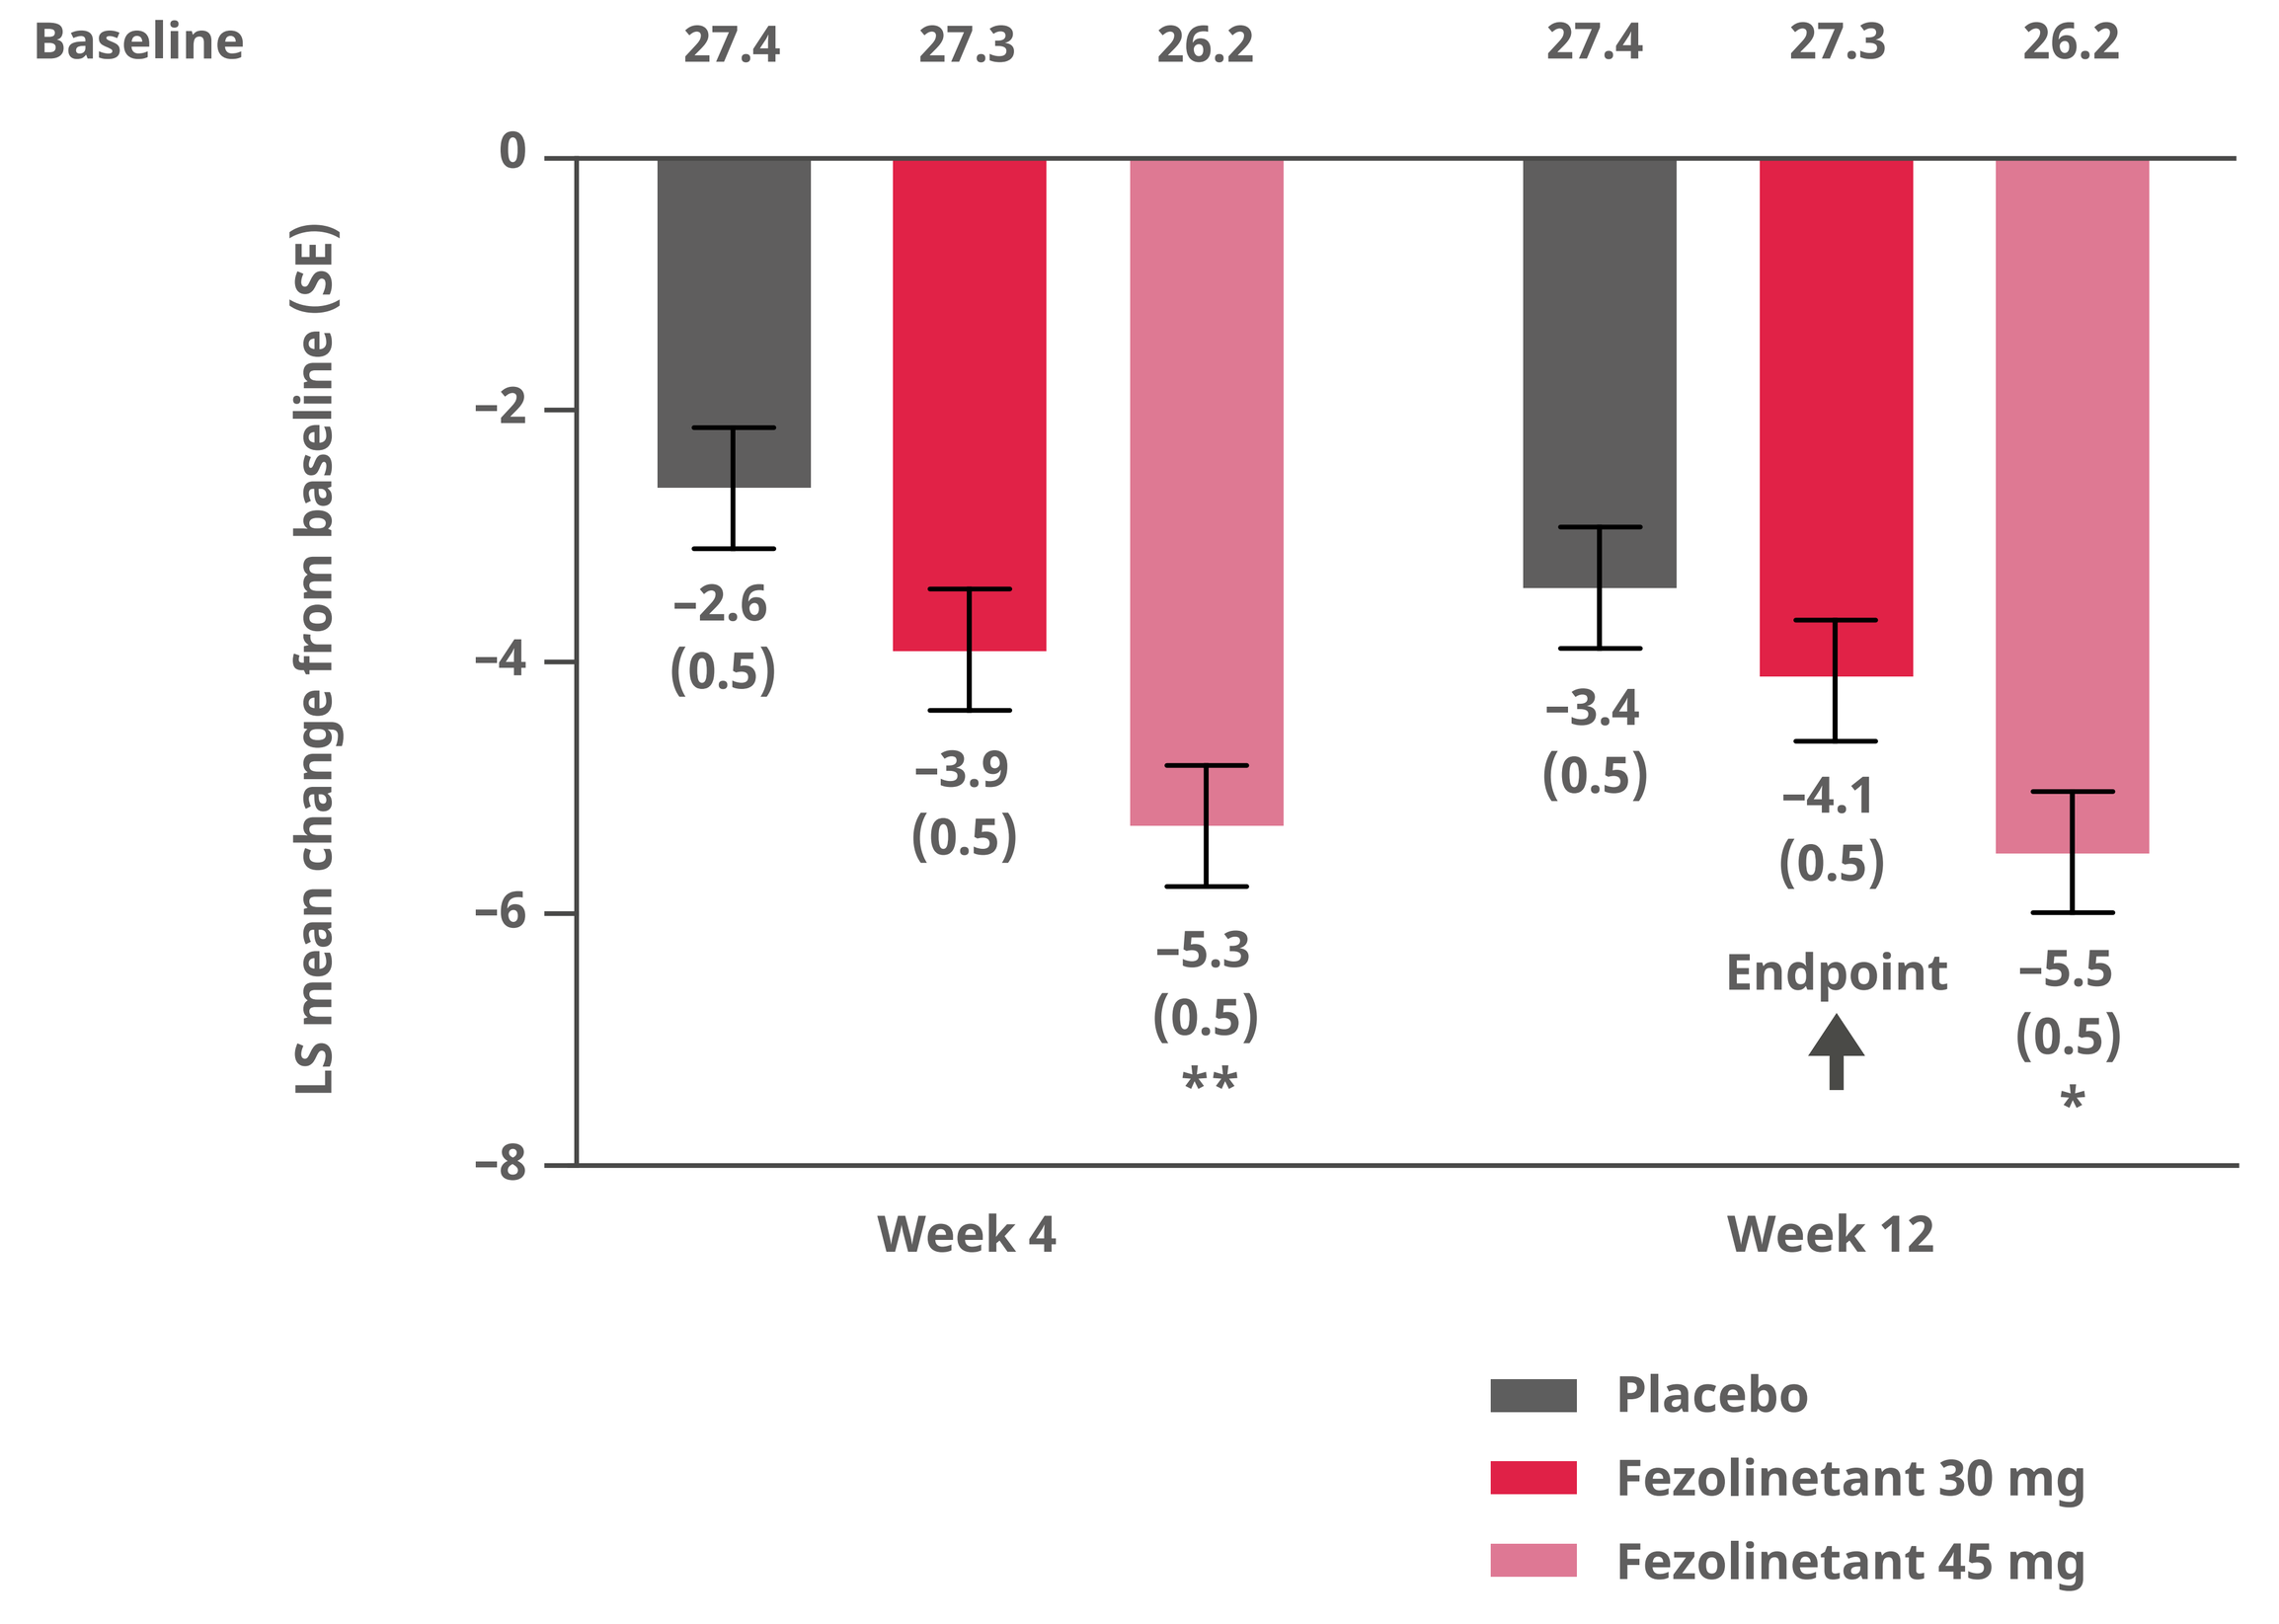
Baseline
27.4 27.3 26.2 27.4 27.3 26.2
LS mean change from baseline (SE)
0
–2
–4
–6
–8
–2.6 (0.5)
–3.9 (0.5)
–5.3 (0.5) **
–3.4 (0.5)
–4.1 (0.5)
Endpoint
–5.5 (0.5) *
Week 4
Week 12
Placebo
Fezolinetant 30 mg
Fezolinetant 45 mg

LS, least squares; PROMIS SD SF 8b, Patient-Reported Outcomes Measurement Information System Sleep Disturbance–Short Form 8b; SE, standard error

*p<0.05; **p<0.001

CONCLUSIONS

- **Fezolinetant 30 mg and 45 mg once daily were efficacious and well tolerated for the treatment of moderate-to-severe VMS associated with menopause**
- **Full safety results will be obtained on completion of the fezolinetant Phase 3 programme**

ACKNOWLEDGEMENTS

The authors would like to thank the study investigators and staff and all the women with moderate-to-severe VMS who took part in the study. Medical writing support was provided by Sue Cooper, CMPP, of Envision Scientific Solutions, Horsham, UK, and funded by Astellas Pharma Inc.

CONFLICT OF INTEREST

All authors have received support for medical writing. KJ, REN, GN-P, MS, PS, RCT, WW and NS are consultants/advisors for Astellas. ME, CF and ML are employees of Astellas Pharma Inc.

FUNDING SOURCES

This study was funded by Astellas Pharma Inc.

REFERENCES

1. Makara-Studzińśka MT, et al. *Prz Menopauzalny*. 2014;13:203-211.
2. Williams RE, et al. *Maturitas*. 2009;62:153-159.
3. English M, et al. *J Patient Rep Outcomes*. 2021;5:37.

Presented at the Royal College of Obstetricians and Gynaecologists (RCOG) World Congress of the European Society of Gynaecology; 13 – 15 June 2022; London, UK (hybrid in-person and virtual event)