THRIVE-2 Phase 3 Trial of Veligrotug in Chronic Thyroid Eye Disease (TED): Efficacy, Safety, and Quality of Life at 15 Weeks

Jimmy Uddin[1] | Antonio Manuel Garrido-Hermosilla[2] | Michael Schittkowski[3] | Onur Konuk[4] | Anja Eckstein[5] | Adrienne Csutak[6] | Edwina Eade[7] | Patrice Rodien[8] | Vickie Lee[9] | Marco Sales-Sanz[10]| Will Conroy[11] | Abhijit Narvekar[11] on behalf of THRIVE-2 Study Group

[1]Moorfields Eye Hospital, London, UK; [2]Hospital Universitario Virgen Macarena, Universidad de Sevilla, Seville, Spain; [3]Universitätsmedizin Göttingen, Göttingen, Germany; [4]Gazi University Medical School, Dept. of Ophthalmology, Ankara, Turkey; [5]Universitätsmedizin Essen, School of Vision; Essen, Germany; [5]Metavisión Arruzafa, Hospital La Arruzafa, Cordoba, Spain; [6]Dept. of Ophthalmology, University of Pécs Medical School, Pécs, Hungary; [7]University of Sydney, New South Wales, Australia; [8]Centre Hospitalier Universitaire d'Angers, Dept. of Endocrinology, Angers, France; [9]Imperial College Healthcare NHS Trust - Western Eye Hospital, London, UK; [10]Hospital Universitario Ramón y Cajal, Madrid, Spain; [11]Viridian Therapeutics, Inc., Waltham, MA, USA.

KEY TAKEAWAYS

- The THRIVE-2 trial of **veligrotug** is the first global phase 3 RCT of IGF-1R inhibition in chronic TED to show significant improvement not only in proptosis but also in diplopia.
- Improvements in TED symptoms translated to rapid and clinically meaningful improvements in QoL on the GO-QOL scale; improvements were observed as early as 3-6 weeks and reached values well over the clinically meaningful threshold (8 points) at Week 15.
- **Veligrotug** was generally well tolerated, with mostly mild AEs and a low discontinuation rate.
- Results suggest **veligrotug** may become a promising new treatment option in TED.

INTRODUCTION

- TED remains a disease with significant unmet need, especially in the chronic phase. More treatment options are needed to improve convenience, efficacy, and/or safety.
- **Veligrotug**, a full antagonist humanized mAb to IGF-1R, was assessed in THRIVE-2 in chronic TED (onset >15 months, proptosis ≥3 mm above normal, and any CAS). Patients received either 10 mg/kg **veligrotug** or placebo given Q3W for a total of 5 IV infusions.
- Safety and efficacy including proptosis, diplopia, CAS, and GO-QOL were evaluated at Week 15.
 - CAS is a 7-point scale used to assess TED-related inflammatory activity.
 - GO-QOL is a disease-specific questionnaire that assesses QoL in TED.
- See RCOphth ePoster for THRIVE results in active TED.

BASELINE CHARACTERISTICS

	Veligrotug (n=125)	Placebo (n=63)
Age, mean years (SD)	50.5 (13.5)	50.7 (12.0)
Female, % (n)	76% (95)	73% (46)
Proptosis (Hertel), mean mm (SD)	24.3 (3.3)	23.8 (3.3)
CAS, mean (SD)	2.7 (1.9)	2.5 (1.8)
CAS 0 or 1, % (n)	35% (44)	35% (22)
CAS ≥3, % (n)	57% (71)	52% (33)
Diplopia at baseline, % (n)	52% (65)	59% (37)
Diplopia, mean Gorman score (SD)[1]	2.0 (0.8)	2.1 (0.9)
Time since onset of TED signs/symptoms, mean months (SD)	69.8 (78.9)	81.7 (83.7)
GO-QOL, mean overall score (visual function + appearance)	61.0	66.8
GO-QOL, mean visual function subscale score	77.1	77.2
GO-QOL, mean appearance subscale score	51.3	56.6

[1]In patients with diplopia at baseline.

EFFICACY AND SAFETY AT WEEK 15

Efficacy endpoints		Veligrotug (n=125)	Placebo (n=63)	p-value
Overall Response	Overall responder rate (MRI/CT)[1]	48%	3%	p < 0.0001
	Overall responder rate (Hertel)	56%	7%	p < 0.0001
Proptosis (Hertel)	Proptosis responder rate[2]	56%	8%	p < 0.0001
	Proptosis mean CFB, mm	-2.34	-0.46	p < 0.0001
Proptosis (MRI/CT)	Proptosis responder rate	48%	3%	p < 0.0001
	Proptosis mean CFB, mm	-2.07	-0.36	p < 0.0001
Diplopia	Diplopia complete resolution[3]	32%	14%	p = 0.0152
	Diplopia responder rate[4]	56%	25%	p = 0.0006
CAS *(prespecified exploratory endpoints)*	CAS 0 or 1[5]	54%	24%	p = 0.0060
	CAS mean CFB[5]	-2.9	-1.3	p < 0.0001

Endpoints shown are primary or secondary endpoints in at least one region, unless otherwise noted. Missing values were imputed using prespecified imputation methodology. Results were obtained from a GEE model with an unstructured covariance matrix. [1]Primary endpoint in Europe and Australia; percentage of participants with ≥2-mm reduction in proptosis (by MRI/CT) AND no worsening in CAS from baseline in the study eye, without corresponding deterioration (≥2-mm/point increase) in proptosis or CAS in the fellow eye of 0 at Week 15. [2]Primary endpoint in North America: percentage of participants with ≥2-mm reduction in proptosis from baseline in the study eye (by Hertel), without deterioration in the fellow eye (≥2-mm increase). [3]Percentage of participants reporting baseline diplopia (Gorman score >0) and a score of 0 at Week 15; [4]Percentage of participants reporting a score reduction of at least 1 on the Gorman subjective diplopia scale, among patients with diplopia at baseline. [5]Of participants with CAS ≥3 at baseline (n=71, veligrotug; n=33, placebo); CAS subpopulation analyses were prespecified exploratory endpoints and statistical p values are for descriptive purposes only.

AEs occurring at ≥10% frequency in either arm Patients, % (n)	Veligrotug (n=125)	Placebo (n=63)
Muscle spasms	36% (45)	6% (4)
Headache	14% (18)	13% (8)
Hearing impairment[1]	13% (16)	3% (2)
Fatigue[1]	12% (15)	8% (5)
Diarrhea	11% (14)	10% (6)
Hyperglycemia[1]	10% (13)	5% (3)
Menstrual disorders[1,2]	33% (16/48)	10% (2/20)

Three serious AEs in 3 **veligrotug** patients: Grade 3 vertigo (related), Grade 2 arthralgia (unrelated), Grade 2 metabolic encephalopathy (unrelated). Two serious AEs in 2 placebo patients: Grade 3 urticaria (related), Grade 3 fatigue (unrelated).
[1]Includes multiple terms aggregated using standard sets of MedDRA terms. [2]Reported as percentage of menstruating women.

- Most AEs were mild in both arms: 75% vs 73% for **veligrotug** vs placebo.
- There was a low treatment discontinuation rate of 6% in the **veligrotug** arm.
- Serious AEs occurred in 5 patients: 3 **veligrotug** and 2 placebo (1 treatment-related per arm).

GO-QOL OUTCOMES*

Overall

(comprised of 2 subscales, with 8 questions each [scale 0-100])

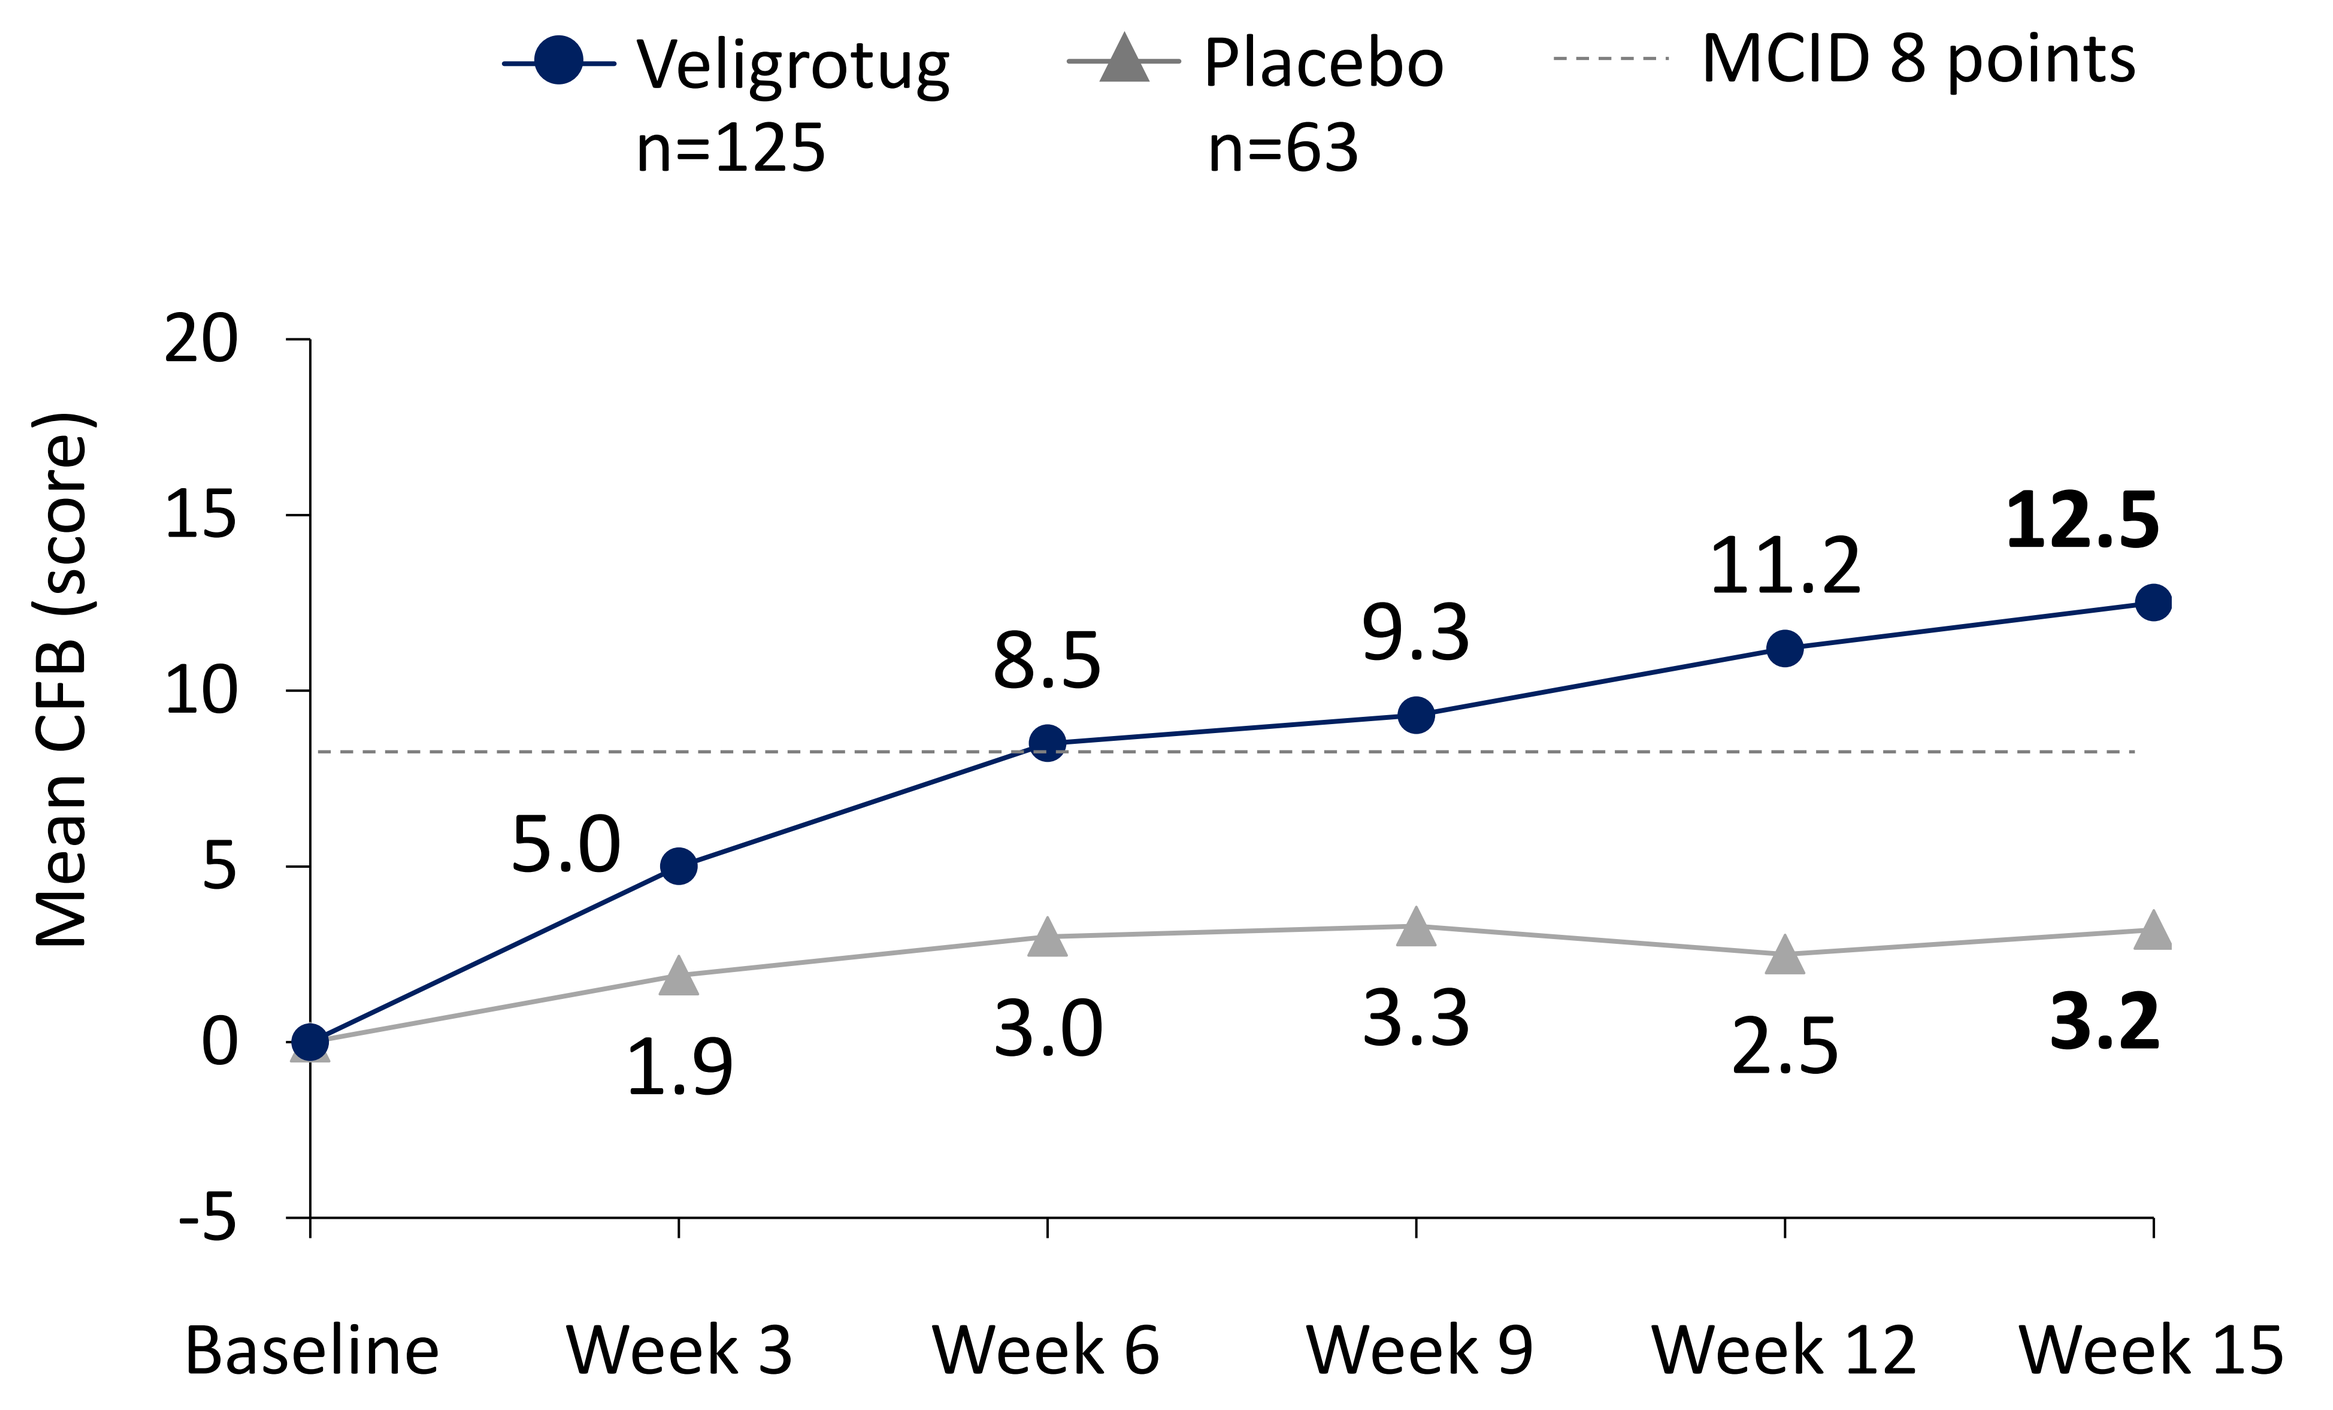
Veligrotug n=125
Placebo n=63
MCID 8 points
Mean CFB (score)
20
15
10
5
0
-5
Baseline
Week 3
Week 6
Week 9
Week 12
Week 15
5.0
8.5
9.3
11.2
12.5
1.9
3.0
3.3
2.5
3.2

Visual function subscale

(limitations in activities: biking, walking, driving, reading, watching TV, moving around house, and hobbies)

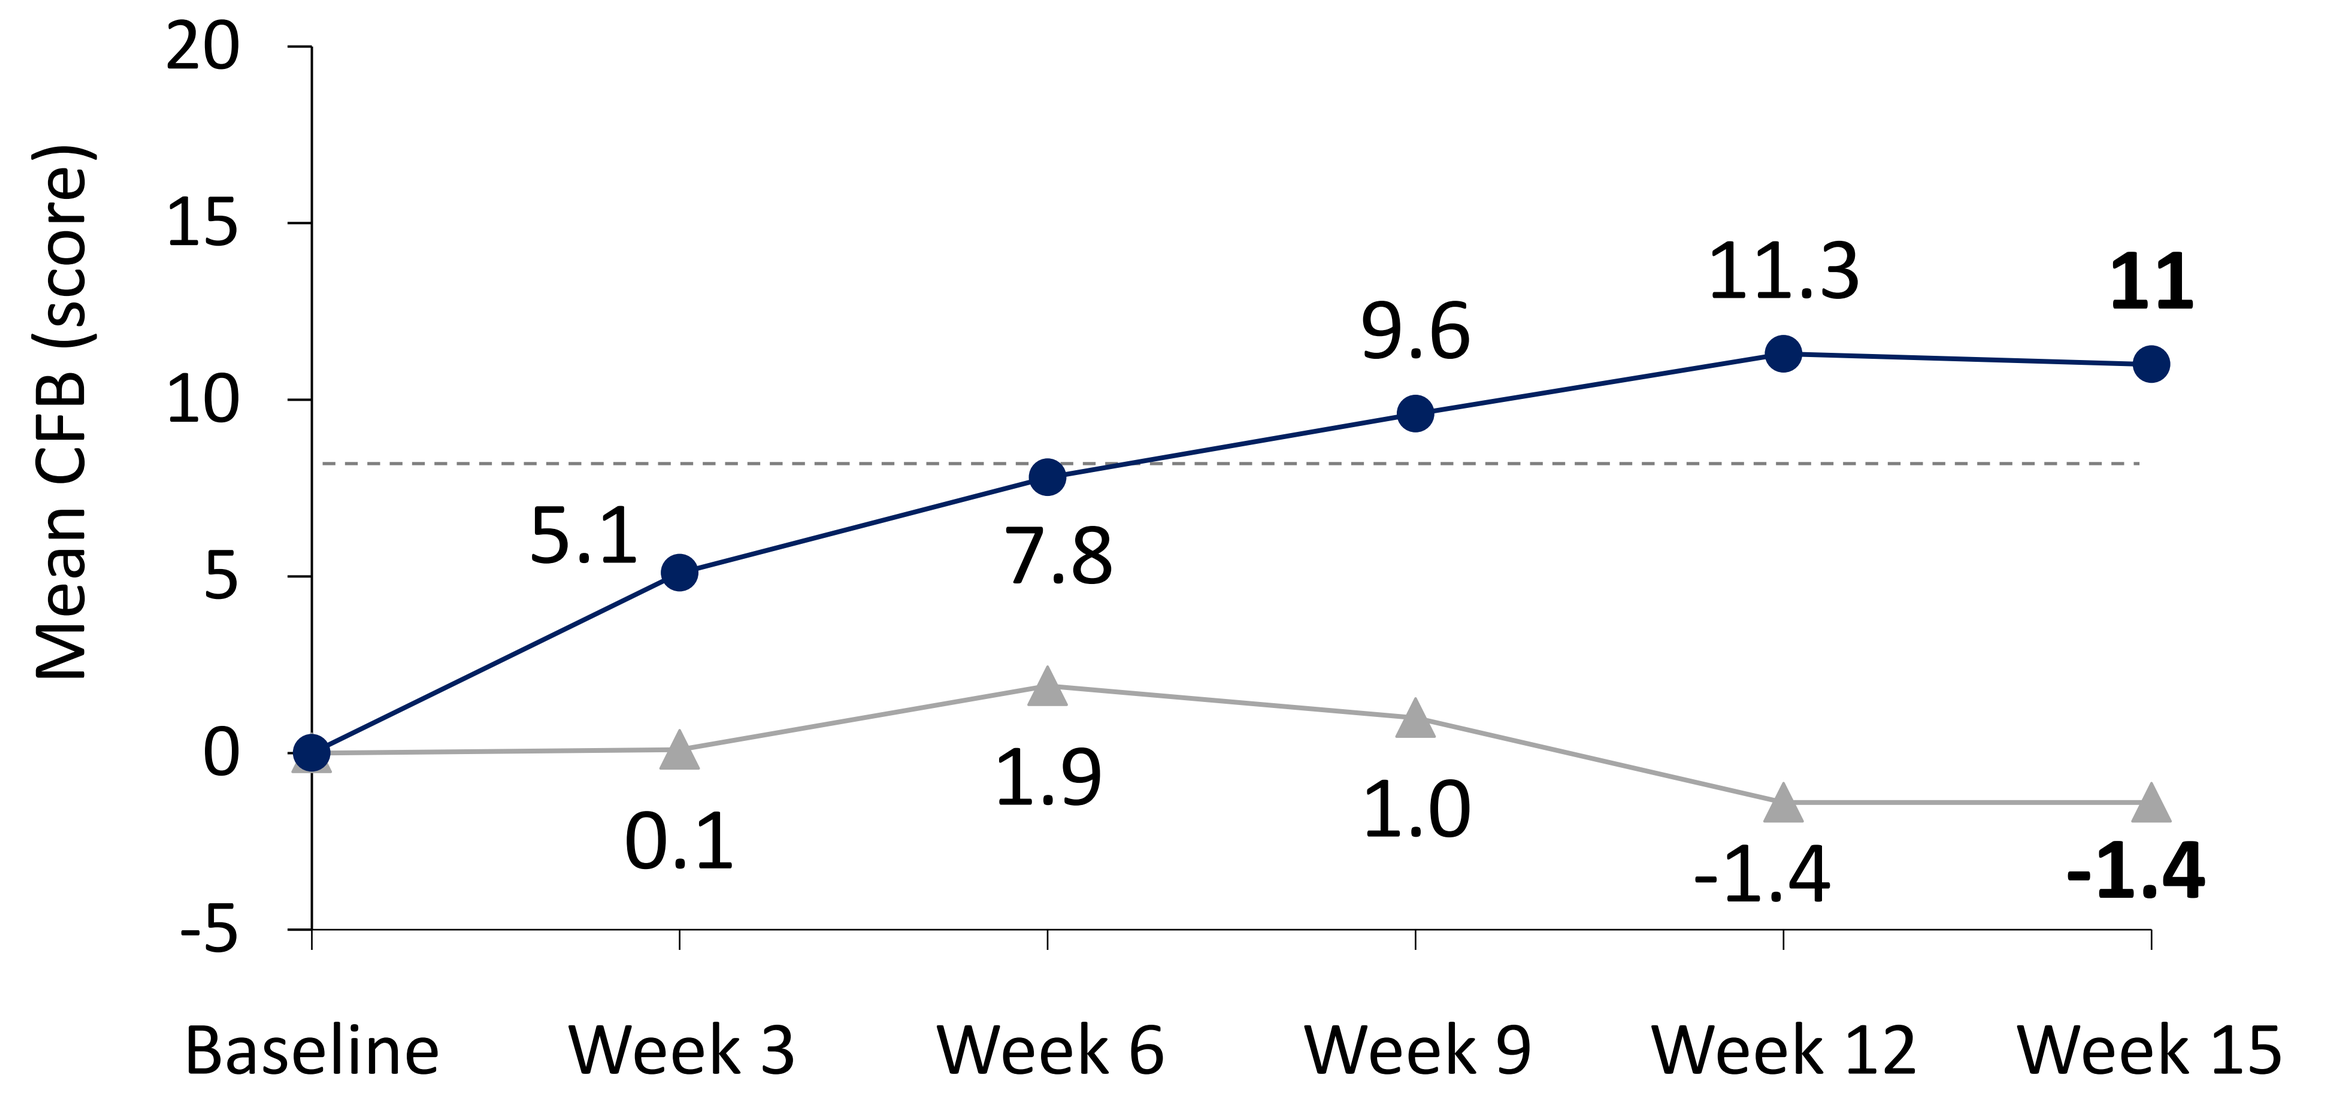
Mean CFB (score)
20
15
10
5
0
-5
Baseline
Week 3
Week 6
Week 9
Week 12
Week 15
5.1
7.8
9.6
11.3
11
0.1
1.9
1.0
-1.4
-1.4

Appearance subscale

(psychosocial influence: changed appearance, reactions from others, self-confidence, social activities, and relationships)

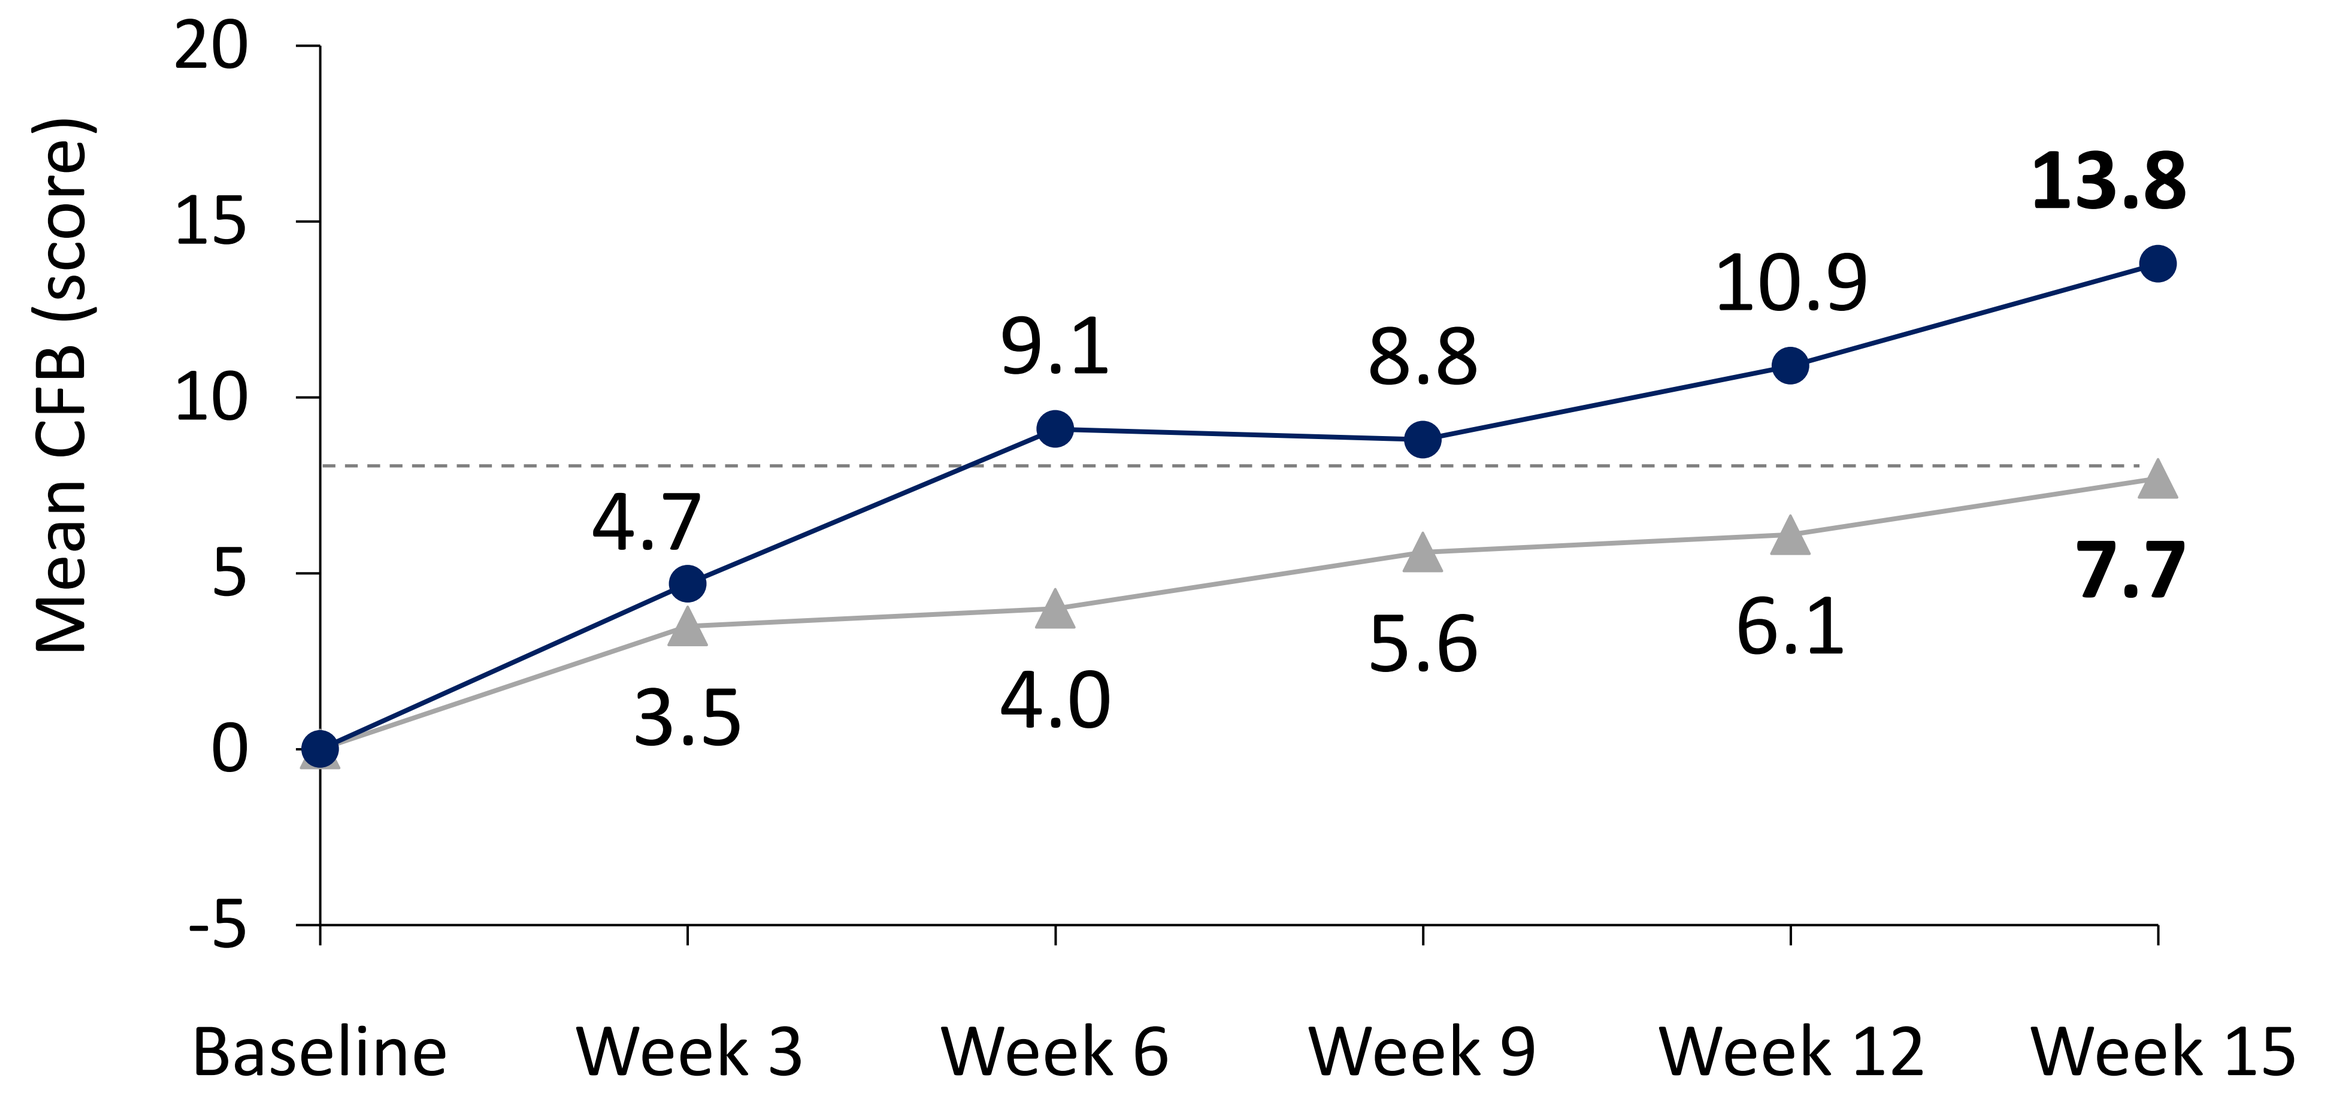
Mean CFB (score)
20
15
10
5
0
-5
Baseline
Week 3
Week 6
Week 9
Week 12
Week 15
4.7
9.1
8.8
10.9
13.8
3.5
4.0
5.6
6.1
7.7

*Prespecified exploratory analysis at Week 15 and post hoc at earlier timepoints. GO-QOL MCID (minimal clinically important difference) vs baseline is 8 points. Nominal p <0.01 for all at Week 15.

Disclosures: Study sponsor: Viridian Therapeutics, Inc. **Veligrotug** (VRDN-001) is investigational only; it's safety and efficacy have not been established by any regulatory authority. Formatting and editorial assistance was provided by Keira Kim, MS, and funded by Viridian Therapeutics, Inc. All authors met the ICMJE authorship criteria and had full access to relevant data. JU, AMGH, MS, OK, AE, AC, EE, PR, and VL are study investigators for Viridian Therapeutics, Inc. WC and AN, are employees of Viridian Therapeutics, Inc. The authors would like to thank the study investigators, research teams, and study participants who made this research possible.

PDF of poster and additional information including THRIVE Study Group: Scan QR code.

Abbreviations used in poster: AE, adverse event; CAS, clinical activity score; CFB, change from baseline; GEE, generalized estimating equations; GO-QOL, Graves' Ophthalmopathy Quality of Life questionnaire; IGF-1R, insulin-like growth factor-1 receptor; IV, intravenous; mAb, monoclonal antibody; MedDRA, Medical Dictionary for Regulatory Activities; SD, standard deviation; QoL, quality of life; Q3W, every 3 weeks; TED, thyroid eye disease.

Trial Information: THRIVE-2, NCT06021054

Contact Information: MedInfo@viridiantherapeutics.com

VIRIDIAN

The Royal College of Ophthalmologists (RCOphth), Manchester, UK; May 18-21, 2026