Congenital Simple Hamartoma of the Retinal Pigment Epithelium (CSHRPE) : a Systematic Review

REDA GHAZALI ; TOBIN SMITH ; SAMIA V. GHAZALI ; MANOLITO L. FINGER

FOR ANY QUERIES : REDAGHAZALI@OUTLOOK.FR

01 Introduction

> **CSHRPE is a small, benign, densely pigmented, nodular macular tumor of the RPE ophthalmologists should learn to recognize.**

Ocular oncology is a source of clinical hassle in daily practice: the risk of overlooking a sight- or life-threatening lesion and, conversely, the risk of overdiagnosis leading to unnecessary investigations is shared by many clinicians. Beyond classically recognized entities, rarer pigmented lesions of the posterior pole remain difficult to characterize, particularly as atypical presentations can act as masqueraders.

First reported by Laqua in 1981 and better characterized since the advent of OCT, CSHRPE is located in the macular region, most often asymptomatic and discovered incidentally on routine fundus examination.

The aim of this first-of-its-kind systematic review is to gather findings from the existing literature on CSHRPE, refine its clinical and multimodal imaging characterization, and improve diagnostic reach in clinical practice.

02 Methods

Systematic review following PRISMA guidelines. PubMed, Scopus, and Web of Science were searched without date or language restrictions, using an extensive string searche : “congenital simple hamartoma of the retinal pigment epithelium”, “CSHRPE”, and related synonyms. Only peer-reviewed publications were deemed eligible; grey literature, conference abstracts, and retracted articles were excluded.

04 Results

44	67	71	2/3
STUDIES	PATIENTS	EYES	CASES INCIDENTAL FINDING

44 studies were included (35 case reports, 79.5% and 9 case series, 20.5%). 67 patients (71 eyes) were pooled, with a slight female predominance (39/67 patients, 58.2%) and a mean age of 35.2 ± 21.3 years (range 6-80; age documented in 63 patients); 33.3% of patients were aged under 18 and only 6.3% over 65. Disease was unilateral in 94.0% of patients (63/67) and CSHRPE presented as multifocal insults in 5 of 71 eyes (7.0%). Lesions were predominantly extrafoveal macular (42 eyes, 59.2%) or foveolar (24 eyes, 33.8%); notably, no lesion was described in the peripheral retina. Discovery was incidental in nearly 2/3 of patients (42/67, 62.7%).

Lesions were consistently described as darkly pigmented (95.8%), nodular (80.3%), elevated (81.7%) or well-circumscribed (74.6%). Classical imaging features were highly consistent across modalities — hyperreflective intraretinal mass with abrupt margins and complete posterior shadowing on OCT, early hypofluorescence with sometimes late peri-lesional staining on FA, and intrinsic vascularization at the superficial capillary plexus on OCTA.

Follow-up was documented for 30/71 eyes (42.3%) with a mean of 39.5 months (median 13, range 0-180). Lesions were stable in 23/30 eyes (77%) and growth was reported in 1 eye only. When described, VA was stable in 16 eyes, improved in 5 eyes and worsened in 1 eye only (among the cases for which progression was reported) . 6 eyes (8.5%) required intervention for vision-threatening complications — macular edema (8.5%) and vitreomacular traction (7.0%) being the most frequent. However, in most cases, a single and solitary lesion was described without associated ocular abnormalities.

03 Multimodal Imaging Atlas

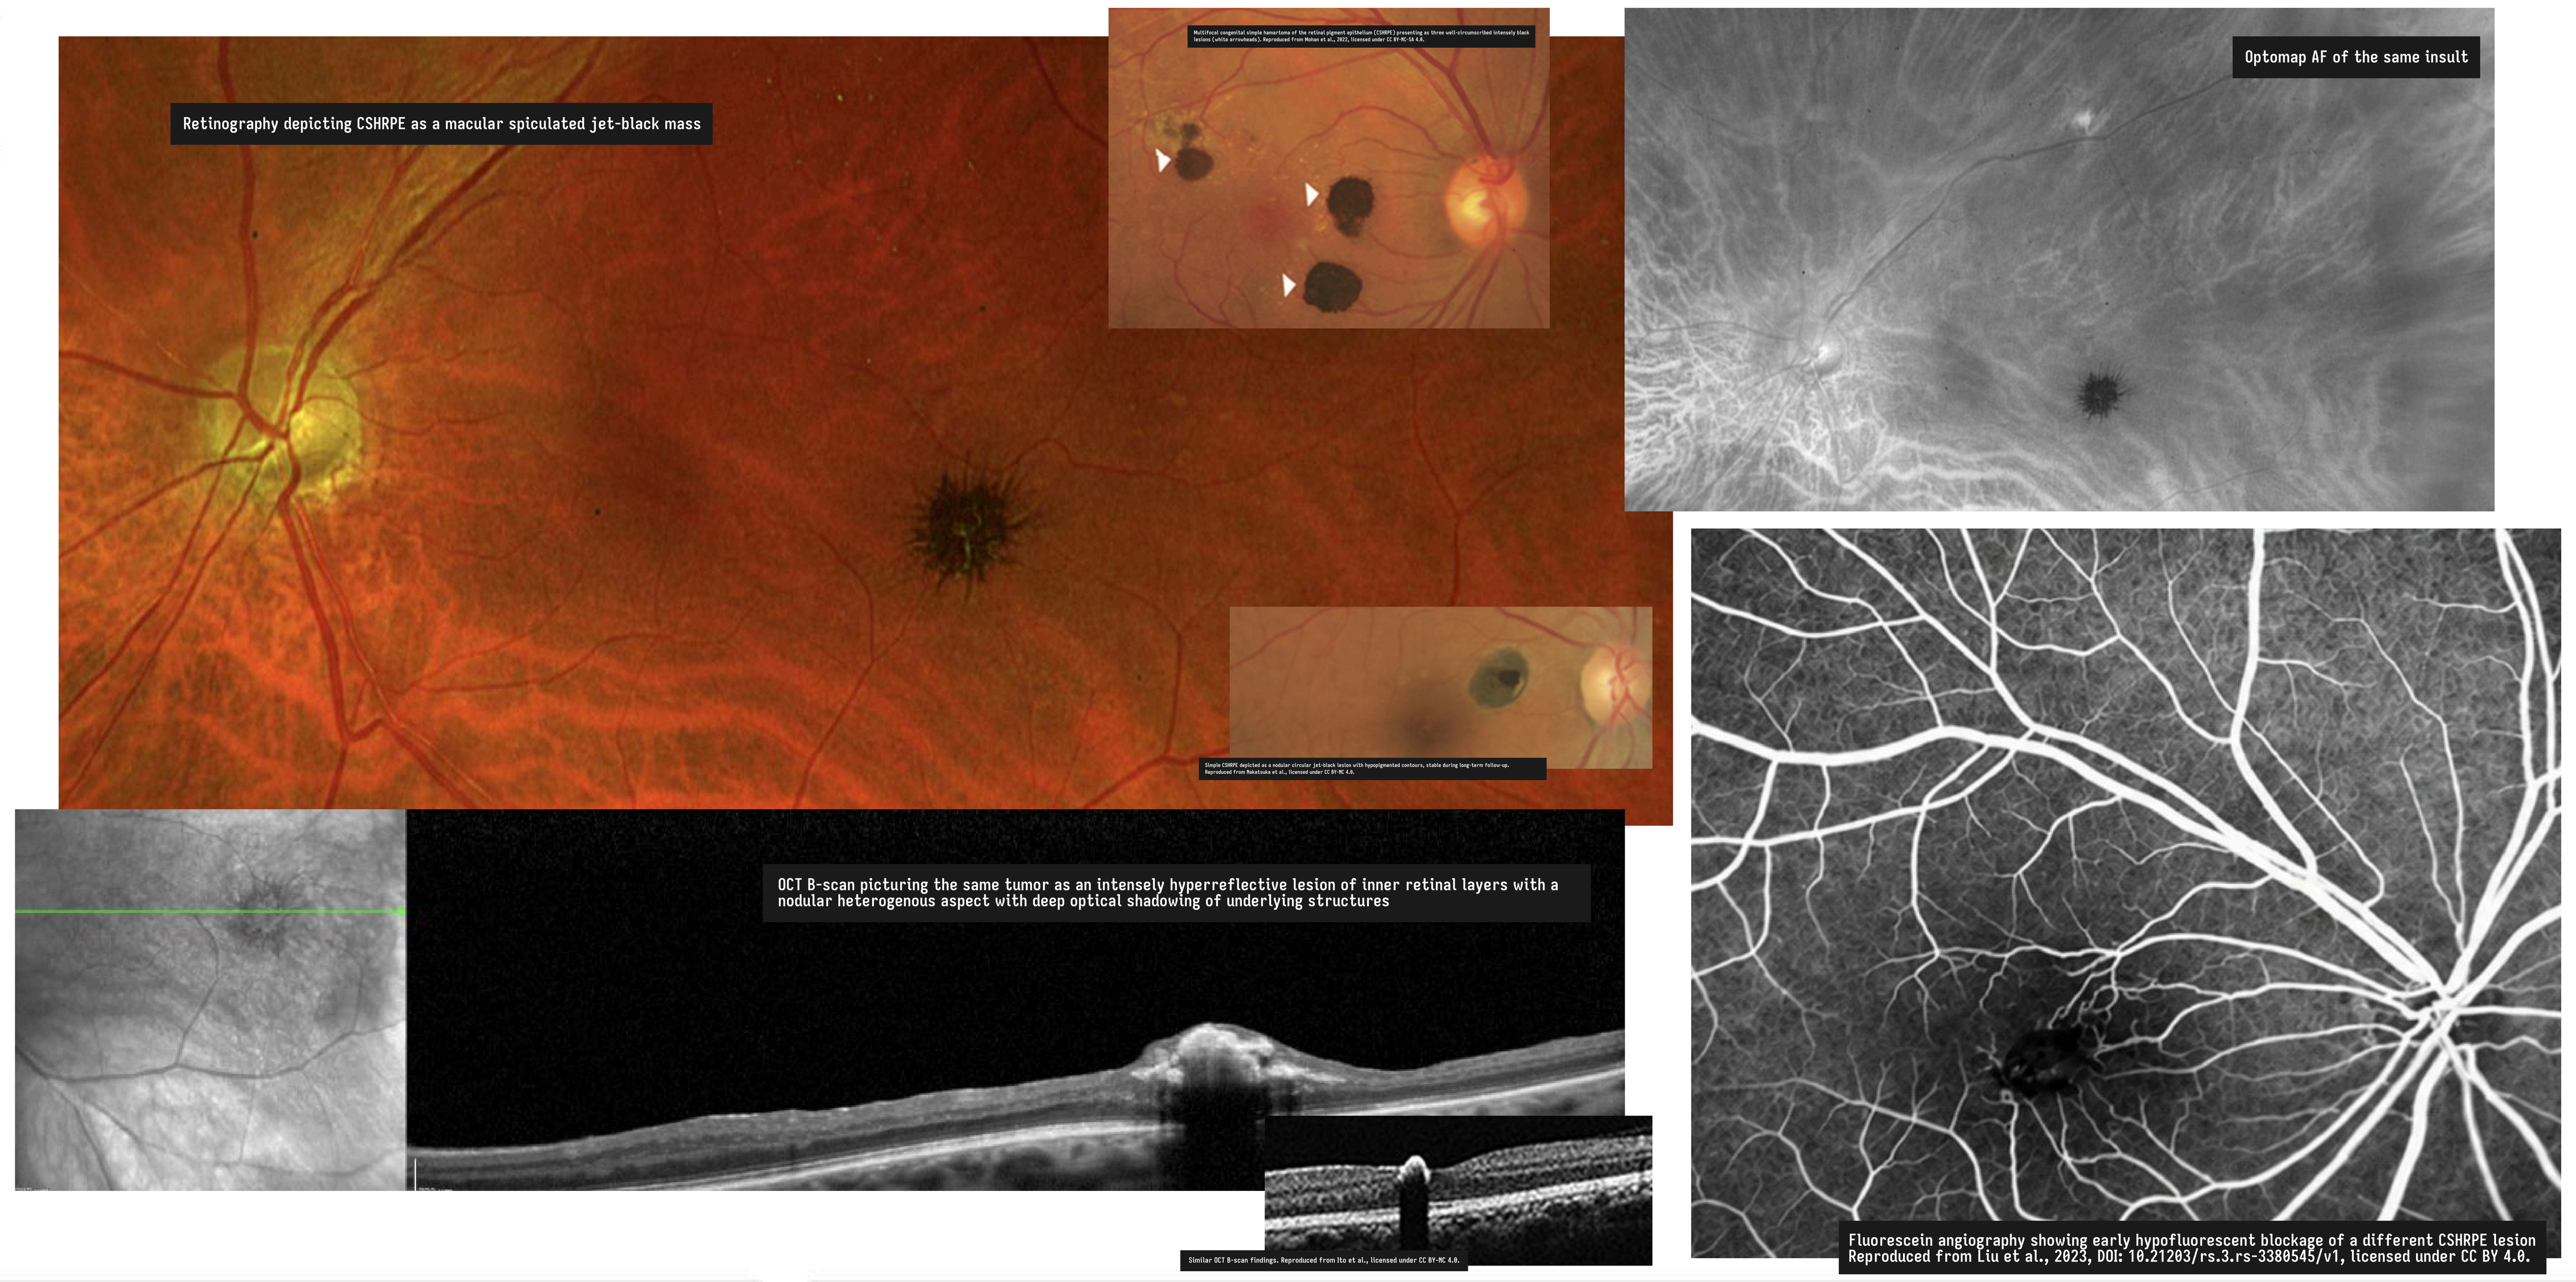
Retinography depicting CSHRPE as a macular spiculated jet-black mass
Optomap AF of the same insult
OCT B-scan picturing the same tumor as an intensely hyperreflective lesion of inner retinal layers with a nodular heterogenous aspect with deep optical shadowing of underlying structures
Fluorescein angiography showing early hypofluorescent blockage of a different CSHRPE lesion
Reproduced from Liu et al., 2023, DOI: 10.21203/rs.3.rs-3380545/v1, licensed under CC BY 4.0.

Included Studies

CHARACTERISTIC	N (%)
Total studies	44 (100.0)
STUDY TYPE	
Case report	35 (79.5)
Case series	9 (20.5)
PATIENTS PER STUDY	
1 patient	35 (79.5)
2 patients	3 (6.8)
3 patients	4 (9.1)
≥ 4 patients	2 (4.5)
PUBLICATION PERIOD	
Before 1990	2 (4.5)
1990 - 1999	0 (0.0)
2000 - 2009	10 (22.7)
2010 - 2019	20 (45.5)
≥ 2020	12 (27.3)

Patient Demographics & Disease Characteristics

CHARACTERISTIC	N (%)
COHORT SIZE	
Number of patients	67
Number of eyes	71
SEX (N = 67 PATIENTS)	
Female	39 (58.2)
Male	24 (35.8)
Not reported	4 (6.0)
AGE, YEARS (N = 63 PATIENTS)	
Mean (SD)	35.2 (21.3)
Median (range)	32 (6-80)
< 18 years	21 (33.3)
≥ 65 years	4 (6.3)
INVOLVEMENT (N = 67 PATIENTS)	
Unilateral	63 (94.0)
Bilateral	4 (6.0)
LATERALITY (N = 71 EYES)	
Right (OD)	40 (56.3)
Left (OS)	27 (38.0)
Not reported	4 (5.6)
LESION LOCATION (N = 71 EYES)	
Extrafoveal macular	42 (59.2)
Foveolar	24 (33.8)
Juxta-/Peripapillary	2 (2.8)
Not reported	3 (4.2)

Clinical & Imaging Characteristics

CHARACTERISTIC	N (%)
MODE OF DISCOVERY (N = 67 PATIENTS)	
Incidental	42 (62.7)
Symptomatic	18 (26.9)
Not reported	7 (10.4)
OPHTHALMIC HISTORY (N = 67 PATIENTS)	
Non-significant / not reported	50 (74.6)
Cataract / pseudophakia	6 (9.0)
Amblyopia / strabismus / esotropia	5 (7.5)
Previous ocular trauma	2 (3.0)
Previous photocoagulation / IVB	2 (3.0)
Optic nerve head anomaly	1 (1.5)
Glaucoma	1 (1.5)
SYSTEMIC HISTORY (N = 67 PATIENTS)	
Non-significant / not reported	58 (86.6)
Diabetes mellitus	5 (7.5)
Sarcoidosis	1 (1.5)
Down's syndrome	1 (1.5)
Congenital heart disease	1 (1.5)
Osteomalacia / hypophosphatemia	1 (1.5)
MORPHOLOGICAL PROPERTIES (N = 71 EYES)	
Darkly pigmented	68 (95.8)
Elevated / protruding	58 (81.7)
Nodular	57 (80.3)
Well-circumscribed	53 (74.6)
Feeder vessels visible	17 (23.9)

Classical Imaging Findings

MODALITY	CLASSICAL FINDINGS
Fundus / Retinophoto	Solitary macular lesion; jet-black; well-circumscribed; nodular; elevated into vitreous; feeder vessels sometimes visible.
OCT	Hyperreflective intraretinal mass with protrusion and vitreous extension; complete posterior shadowing; abrupt margins; surrounding choroid and retina usually preserved but can be associated to traction from the vitreous or macular edema.
FA	Early hypofluorescence; sometimes late mild peri-lesional staining; usually no leakage; sometimes a ring of late hyperfluorescence contouring the lesion; can help visualise feeder vessels when present.
FAF	Hypoautofluorescence at the site of the lesion.
B-scan US	Hyperechoic surface lesion; moderate-to-high internal reflectivity; no posterior acoustic shadowing; no choroidal/scleral invasion; may not detect smaller lesions.
OCTA	Inconsistent; intrinsic vascularization at superficial/inner capillary plexus; can show visible feeder/draining vessels. Few studies available.

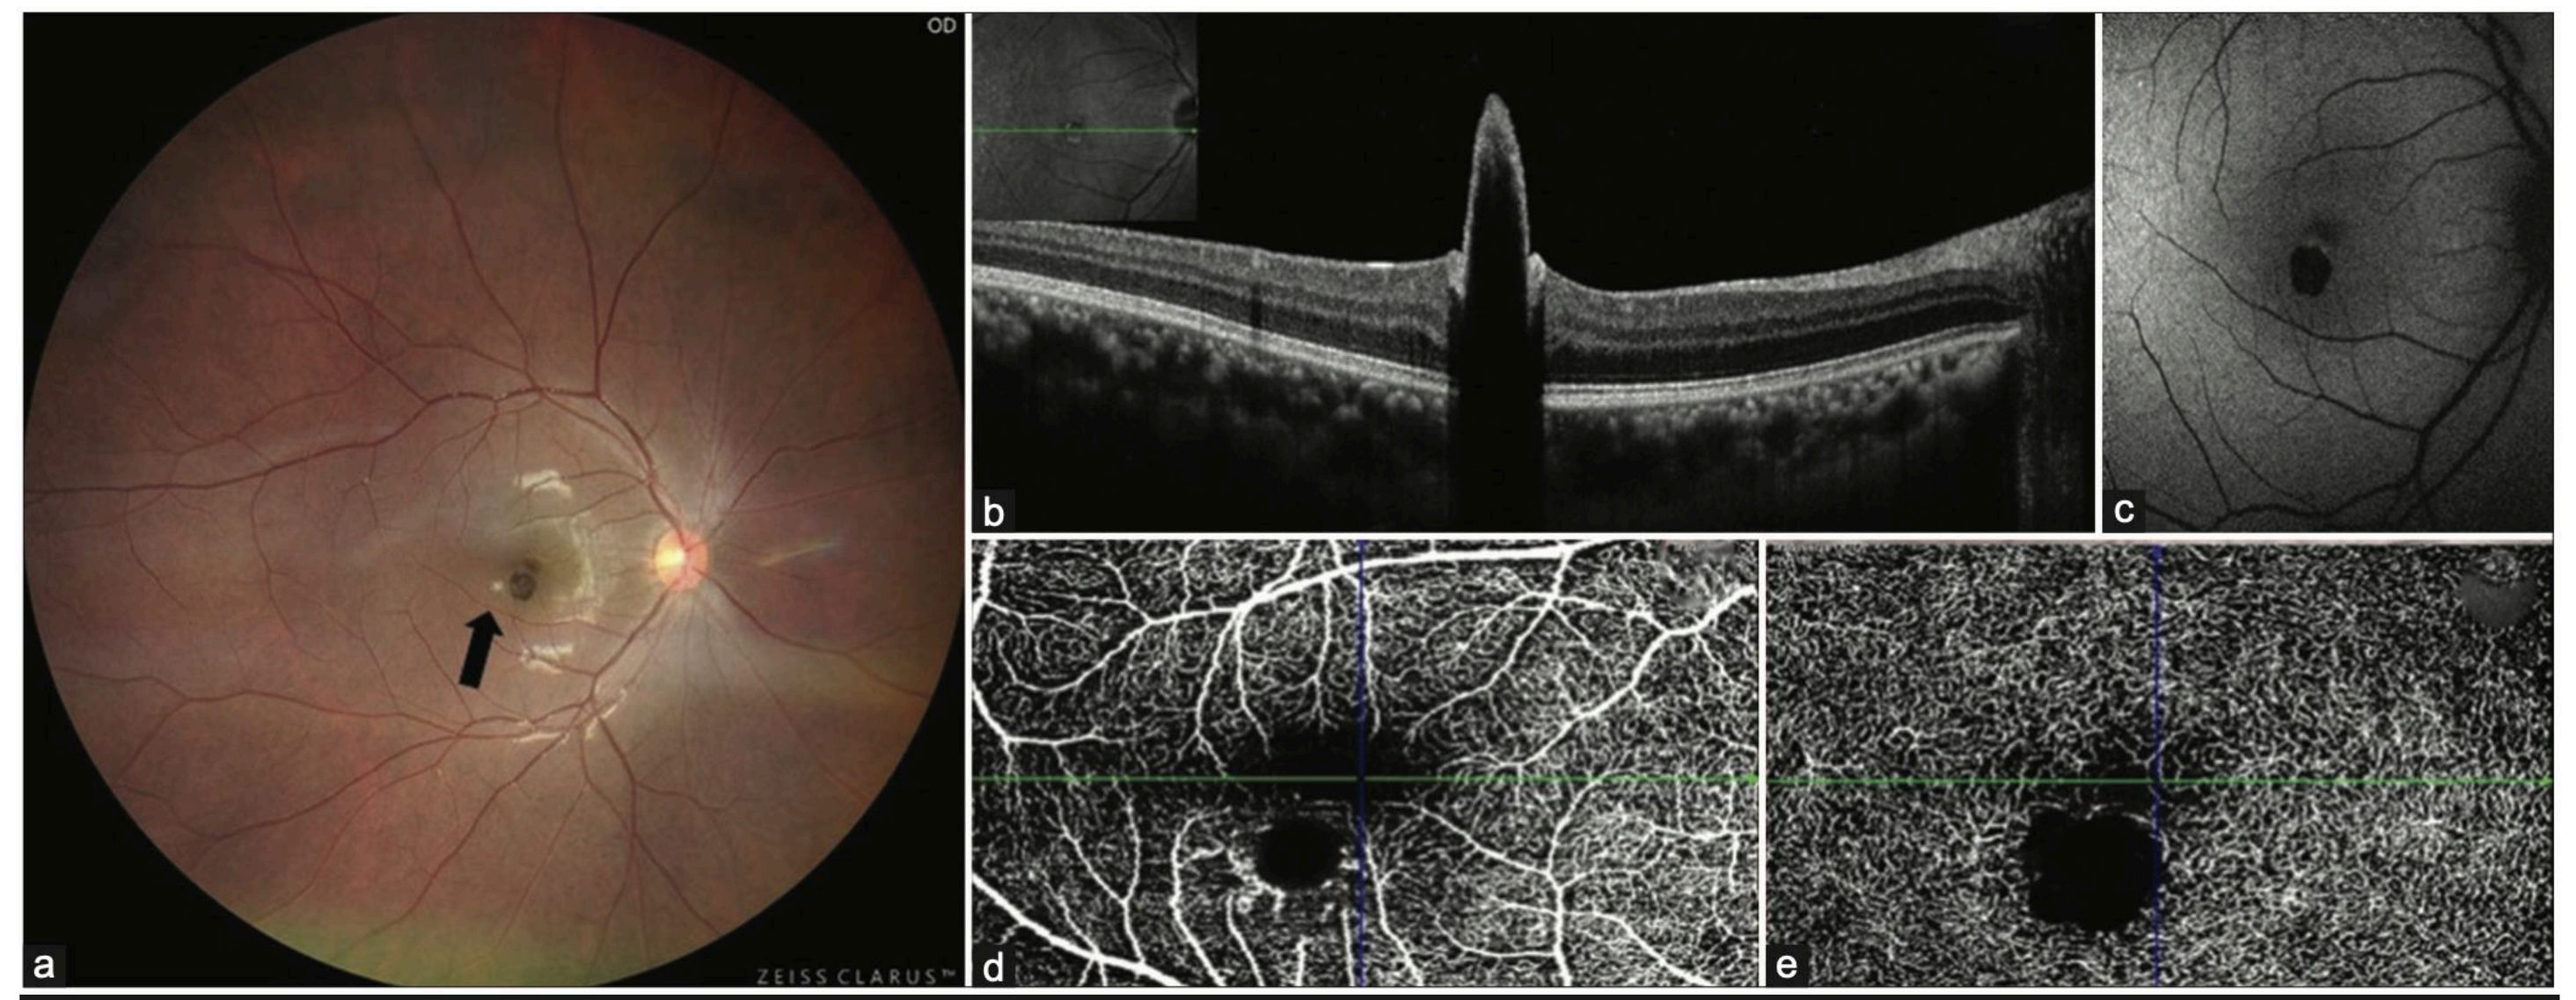
OD
a
b
c
d
e
ZEISS CLARUS

Figure 1. Multimodal imaging of congenital simple hamartoma of the retinal pigment epithelium (CSHRPE). (a) Color fundus photography of the right eye showing a small, intensely black, sharply demarcated subfoveal lesion. (b) Spectral-domain OCT demonstrating a nodular hyperreflective elevation with marked posterior shadowing. (c) Fundus autofluorescence revealing corresponding hypo-autofluorescence. (d,e) OCT-angiography of the superficial and deep capillary plexuses showing blocked flow signals at the level of the lesion with preserved surrounding vasculature. Reproduced from Baghyalakshmi et al., licensed under CC BY-NC-SA 4.0.

05 Conclusion

CSHRPE is a **rare, benign, typically small solitary asymptomatic RPE lesion** most often discovered incidentally, with highly consistent multimodal imaging features and preserved visual function in the vast majority of cases. Complications are infrequent and rarely vision-threatening. Macular predilection is a central point but could also be explained by a lack of peripheral reporting due to low diagnostic reach. **Long-term standardized follow-up with multimodal imaging is needed**, and ocular oncology referral should be considered in atypical or doubtful cases.