The Retinal Hazard of Racket Sports Practice: a Systematic Review

REDA GHAZALI and PROF. ELIE MOTULSKY ·

FOR ANY QUERIES: REDAGHAZALI@OUTLOOK.FR

01 Introduction

Racket sports are popular worldwide and recognized as high-risk for ocular trauma. The full extent of retinal and posterior segment injury attributable to them has never been systematically characterised.

Ocular trauma accounts for a substantial share of emergency consultations, with retinal disorders a leading cause of severe injury — durably threatening prognosis and the social participation of often young patients. Racket sports are practiced by hundreds of millions worldwide and share a common feature: a racket and a high-velocity projectile.

Beyond legacy sports — tennis, squash, badminton — the field has recently been transformed by the explosive growth of **padel and pickleball.** Their safety profile places racket sports within the high-risk category for ocular injury, driven by the kinetic energy of projectiles and, in several disciplines, a projectile diameter close to that of the bony orbital aperture, allowing direct impact on the globe.

This **first-of-its-kind systematic review** aims to define the clinical spectrum, sport distribution, mechanisms, visual outcomes and preventive implications of racket sport-related posterior segment injury, to guide clinical practice and prevention.

02 Methods

Systematic review following **PRISMA guidelines**. Search across **Scopus**, **MEDLINE** (through PubMed), **Web Of Science** and **Cochrane CENTRAL** without language, geographical or sample-size restriction.

Eligible: peer-reviewed publications of any design reporting ≥ 1 posterior-segment lesion sustained during a racket sport (tennis, squash, badminton, padel, pickleball, racquetball, table tennis). Open-globe injuries with vitreous loss/prolapse included. Grey literature, retracted articles, editorials and narrative reviews without patient-level data were excluded.

Methodology based on strict **patient-level extraction**: only data formally specified for the retinal-injury subset were retained; cohort-level proxies were excluded.

04 Results — Key Findings

36 STUDIES	**211** PATIENTS	**357** LESION EVENTS	**0**/120 PROTECTIVE EYEWEAR

36 articles spanning **1973 - 2025** (22 case reports/series, 14 cohort studies) pooled **≥ 211 patients** with racket sport-related posterior segment injury. **Badminton** accounted for the largest share of patients (94, 44.5%), followed by squash (49, 23.2%) and tennis (41, 19.4%); padel (18, 8.5%), racquetball (5, 2.4%), pickleball (3, 1.4%) and table tennis (1, 0.5%) followed. Articles on padel and pickleball appeared only **from 2022 onward**, reflecting the explosive growth of these disciplines.

Event-level reconstruction identified 357 posterior segment lesion events. The commotio retinae was the most frequent posterior segment insult described (103 events, 28.9% of total events), followed by the rhegmatogenous spectrum (86, 24.1% — comprising 58 retinal detachments, 22 tears/breaks/holes, 6 dialyses) and vitreous haemorrhage (55, 15.4%). Choroidal/Bruch's membrane ruptures (29, 8.1%), other intraocular haemorrhages (28, 7.8%) and macular structural lesions (21, 5.9%) completed the spectrum.

Patients were predominantly young adult **males** (4.4 : 1 M : F among the 49 with documented sex), with a median age of 33.5 years (range 11 - 76). **No retinal-injured patient wore sport-specific protective eyewear at impact** (0 / 120 documented).

Direct projectile impact (ball / shuttlecock) was the leading mechanism of injury, driving 70 / 211 insults where information about mechanism was reported.

03 Lesion Spectrum & Sport Distribution

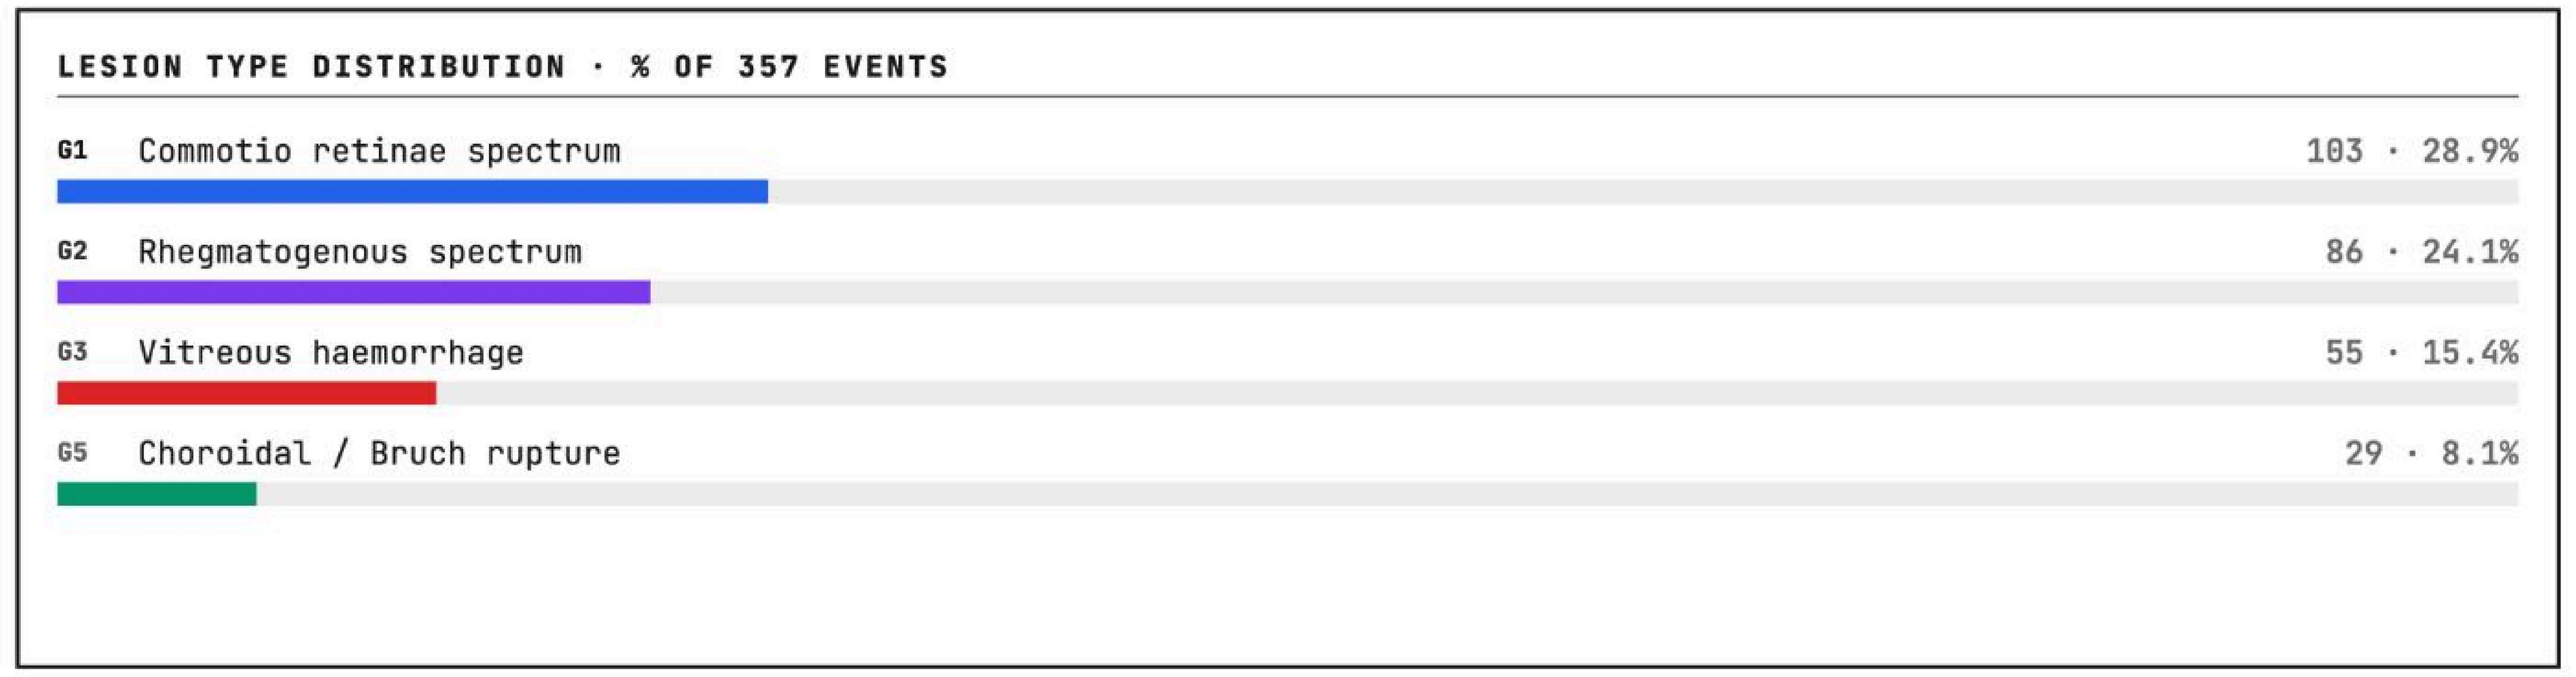
LESION TYPE DISTRIBUTION · % OF 357 EVENTS
G1 Commotio retinae spectrum
103 · 28.9%
G2 Rhegmatogenous spectrum
86 · 24.1%
G3 Vitreous haemorrhage
55 · 15.4%
G5 Choroidal / Bruch rupture
29 · 8.1%

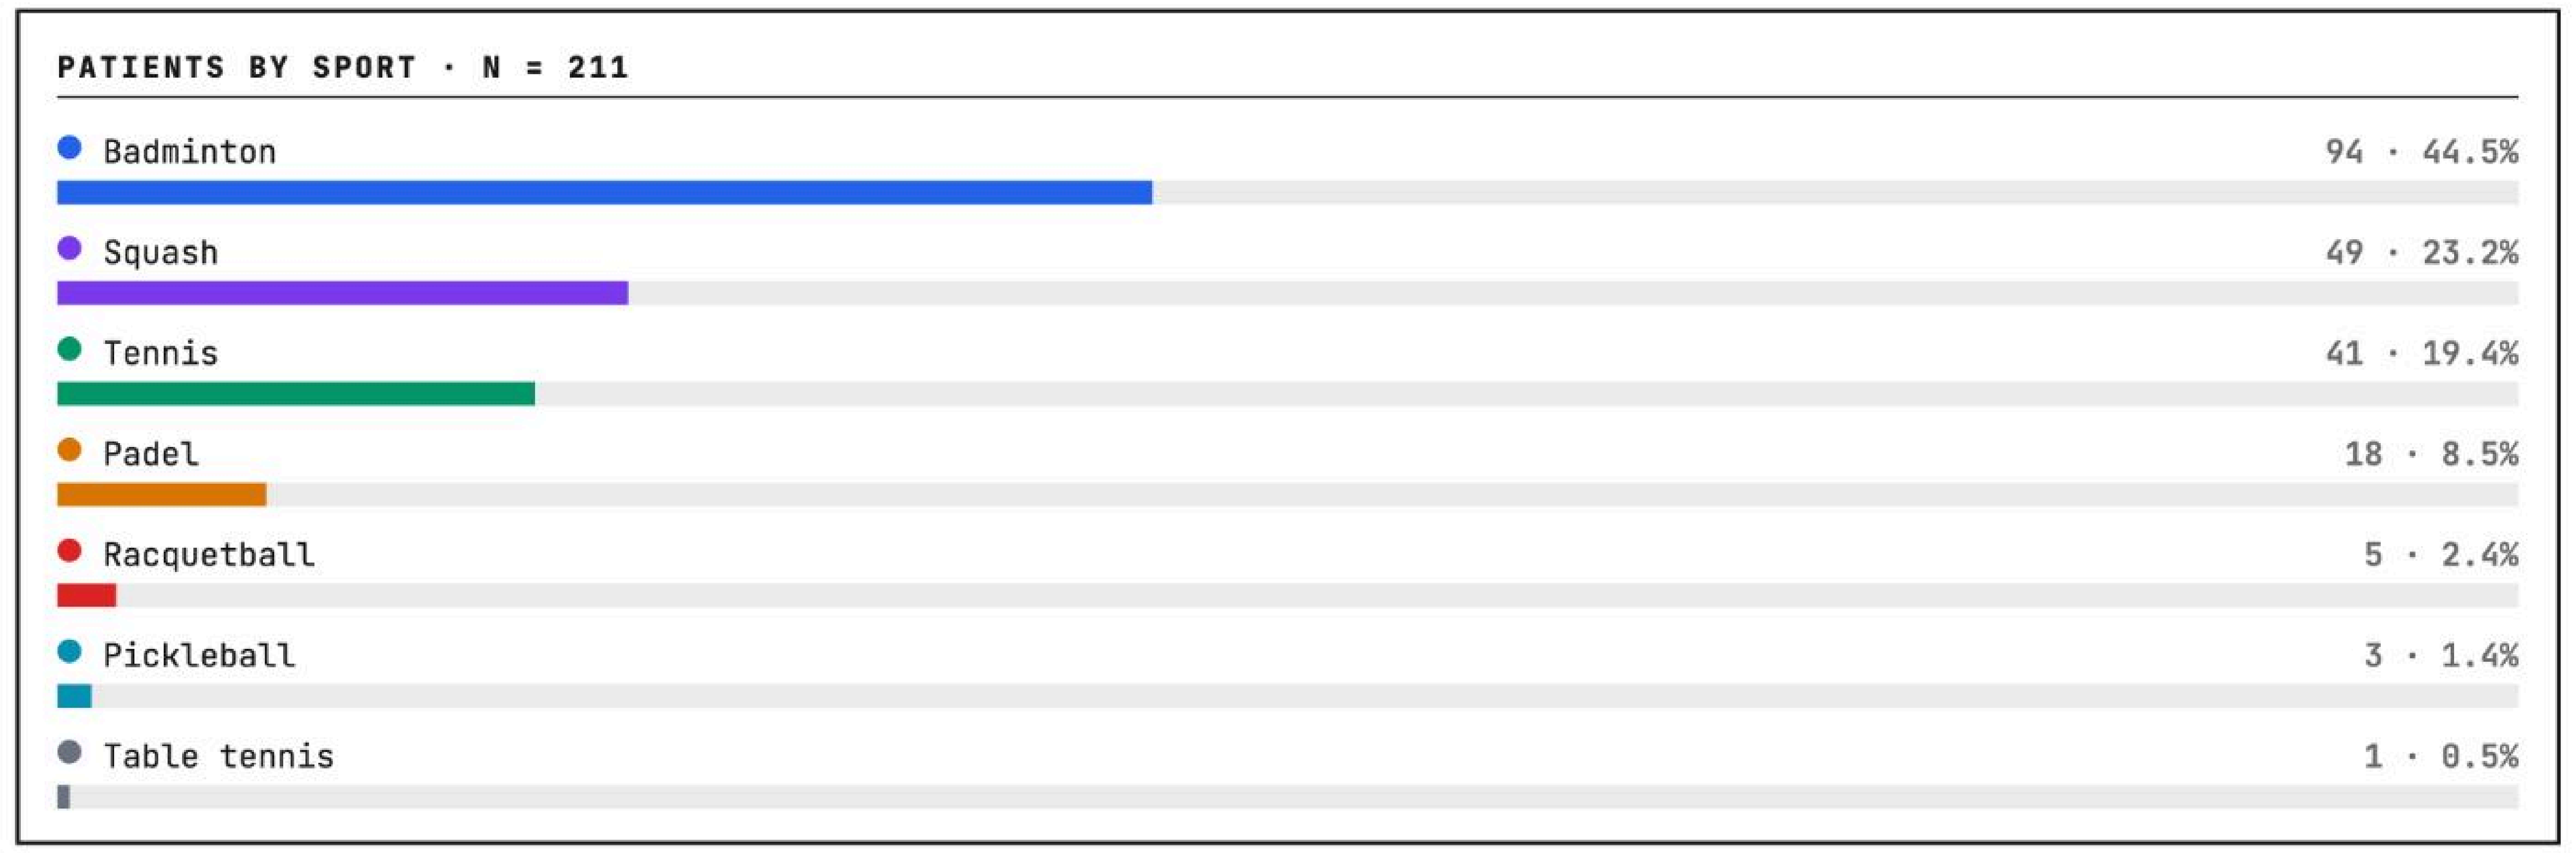
PATIENTS BY SPORT · N = 211
Badminton
94 · 44.5%
Squash
49 · 23.2%
Tennis
41 · 19.4%
Padel
18 · 8.5%
Racquetball
5 · 2.4%
Pickleball
3 · 1.4%
Table tennis
1 · 0.5%

Characteristics of Included Studies

VARIABLE	ARTICLES	PATIENTS
STUDY DESIGN		
Case report	14 (38.9)	14 (6.6)
Case series	8 (22.2)	31 (14.7)
Retrospective cohort	11 (30.6)	147 (69.7)
Prospective cohort	3 (8.3)	19 (9.0)
Total	36 (100)	211 (100)
PUBLICATION PERIOD		
Before 1990	8 (22.2)	62 (29.4)
1990 - 2000	2 (5.6)	27 (12.8)
2000 - 2010	6 (16.7)	8 (3.8)
2010 - 2020	9 (25.0)	23 (10.9)
Since 2020	11 (30.6)	91 (43.1)
SPORT SCOPE		
Single sport	30 (83.3)	–
Multiple sports	6 (16.7)	–
ARTICLES PER SPORT (ANY INVOLVEMENT)		
Tennis	19 (52.8)	–
Badminton	9 (25.0)	–
Squash	7 (19.4)	–
Padel	2 (5.6)	–
Racquetball	2 (5.6)	–
Pickleball	2 (5.6)	–
Table tennis	1 (2.8)	–

Patient Demographics & Sport Distribution

CHARACTERISTIC	N (%)
COHORT TOTALS	
Patients (lower-bound count)	211
Lesion events	357
PATIENTS BY SPORT (% OF 211)	
Badminton	94 (44.5)
Squash	49 (23.2)
Tennis	41 (19.4)
Padel	18 (8.5)
Racquetball	5 (2.4)
Pickleball	3 (1.4)
Table tennis	1 (0.5)
SEX — PATIENT-LEVEL (N = 49 DOCUMENTED)	
Male	40 (81.6)
Female	9 (18.4)
M : F ratio	4.4 : 1
Not individually documented	162 (76.8)
AGE, YEARS (PATIENT-LEVEL DATA)	
Median (range)	33.5 (11 - 76)
18 - 39 yr	21 (48.8)
40 - 59 yr	13 (30.2)
< 18 yr	7 (16.3)
≥ 60 yr	2 (4.7)
LATERALITY (N = 27 DOCUMENTED)	
Left eye (OS)	17 (8.1)
Right eye (OD)	10 (4.7)
Not documented	184 (87.2)

Posterior Segment Lesion Profile

LESION CATEGORY	N (%)
LESION TYPE (% OF 357 EVENTS)	
G1 Commotio retinae spectrum	103 (28.9)
G2 Rhegmatogenous spectrum	86 (24.1)
↳ Retinal detachment	58 (16.2)
↳ Retinal tear / break / hole	22 (6.2)
↳ Retinal dialysis	6 (1.7)
G3 Vitreous haemorrhage	55 (15.4)
G5 Choroidal / Bruch rupture	29 (8.1)
G4 Other intraocular haemorrhage	28 (7.8)
G9 Posterior segment unspecified	23 (6.4)
G6 Macular structural lesions	21 (5.9)
G7 Late-onset complications	7 (2.0)
G8 Optic nerve injury	5 (1.4)
TIMING (% OF 357 EVENTS)	
At presentation	333 (93.3)
Late-onset (at follow-up)	24 (6.7)

Mechanism & Eyewear at Impact

VARIABLE	N (%)
MECHANISM OF INJURY (% OF 211)	
Ball / shuttlecock impact	70 (33.2)
Racket-frame impact	5 (2.4)
Not specified	136 (64.5)
SPORT-SPECIFIC PROTECTIVE EYEWEAR (N = 120 DOCUMENTED ACROSS 14 ARTICLES *)	
Worn	0 (0.0)
Not worn	120 (100.0)
FOOTNOTES	
* Protective eyewear documented in 14 articles (Atkinson, Boopathiraj, Capão Filipe, Chandran, Doxanas, Easterbrook, Gregory, Guo, Ingram, Kasiga, Kawamata, Knorr, MacEwen, Yu).	

05 Conclusion

Racket sport trauma produces a **broad spectrum of posterior segment injury**, from transient commotio retinae to sight-threatening rhegmatogenous and choroidal lesions — sometimes with late onset. **Direct projectile impact** and **absence of certified protective eyewear** are recurrent features across all included disciplines. Comprehensive peripheral retinal examination, appropriate follow-up, screening of late-onset complications, and active promotion of **sport-specific protective eyewear conforming to ASTM F803 or equivalent national standards** should be central to clinical and preventive strategies — particularly in the rapidly growing disciplines of padel and pickleball. We believe a huge share of racket sport injury is preventable with proper measures. Patients at risk should be counselled and informed about this ocular hazard.