From Hopeless to Functional: Multidisciplinary Management of Full-Arch Implant Rehabilitation Using Provisionalization During Staged Procedures

Jen Chen, Harold Baumgarten

Department of Periodontics, University of Pennsylvania School of Dental Medicine, Philadelphia, Pennsylvania, USA

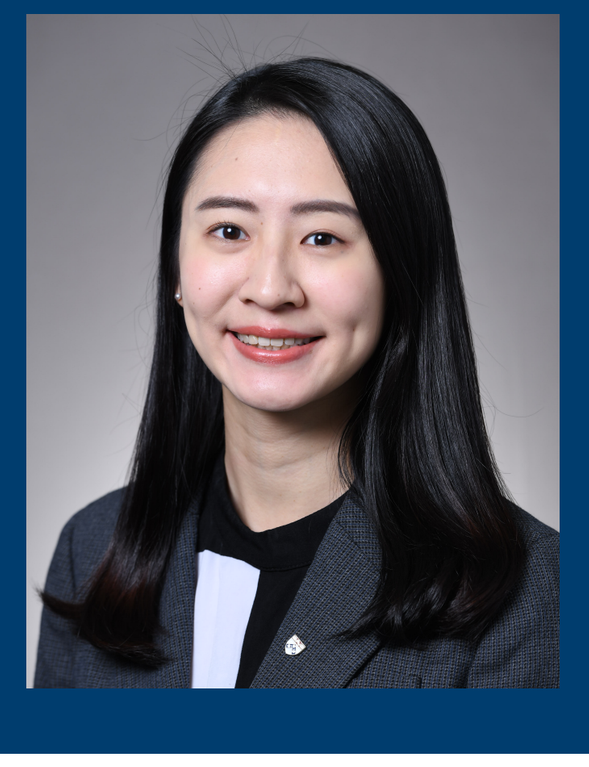

Background

Full-arch implant rehabilitation in cases of advanced bone loss presents significant surgical and prosthetic challenges. Staged extractions combined with guided bone regeneration (GBR) are often necessary to optimize implant placement and long-term outcomes. However, careful planning of the interim restorative phase is equally critical to maintain function, esthetics, and reference points during healing.

Procedure

A 53-year-old female presented with severe periodontitis and hopeless maxillary dentition. Given the extent of bone atrophy, immediate implant placement was not feasible. Staged extractions were performed, initially at sites planned for implant placement, followed by guided bone regeneration (GBR) using particulate allograft and resorbable membranes. The remaining maxillary teeth were planned for extraction after implant placement and loading. To manage existing endodontic lesions and preserve provisional abutment teeth, root canal therapy was performed. A long-span provisional bridge was fabricated in collaboration with a prosthodontist to preserve tooth position references, maintain occlusion, and protect grafted sites during healing. The patient was monitored with 3-month periodontal maintenance visits throughout the healing phase.

Outcomes

This approach preserved vertical dimension, function, and soft tissue contours. Six-month postoperative CBCT confirmed sufficient bone regeneration, enabling the planned placement of seven implants in ideal positions. To accommodate native bone angulation, Southern Co-Axis 12° angle-correction implants were selected for the anterior and premolar regions. Due to ridge height discrepancy between the left and right maxilla, alveoloplasty was planned on the upper left side to align implant platforms. The patient is now transitioning to the final implant placement and prosthetic phase, with implant surgery scheduled for November.

Conclusions

This case demonstrates the effectiveness of a multidisciplinary staged extraction and GBR approach, combined with provisionalization, in managing severe bone loss for full-arch implant rehabilitation. Maintaining dental references and function during healing can contribute to more accurate implant placement and better prosthetic outcomes.

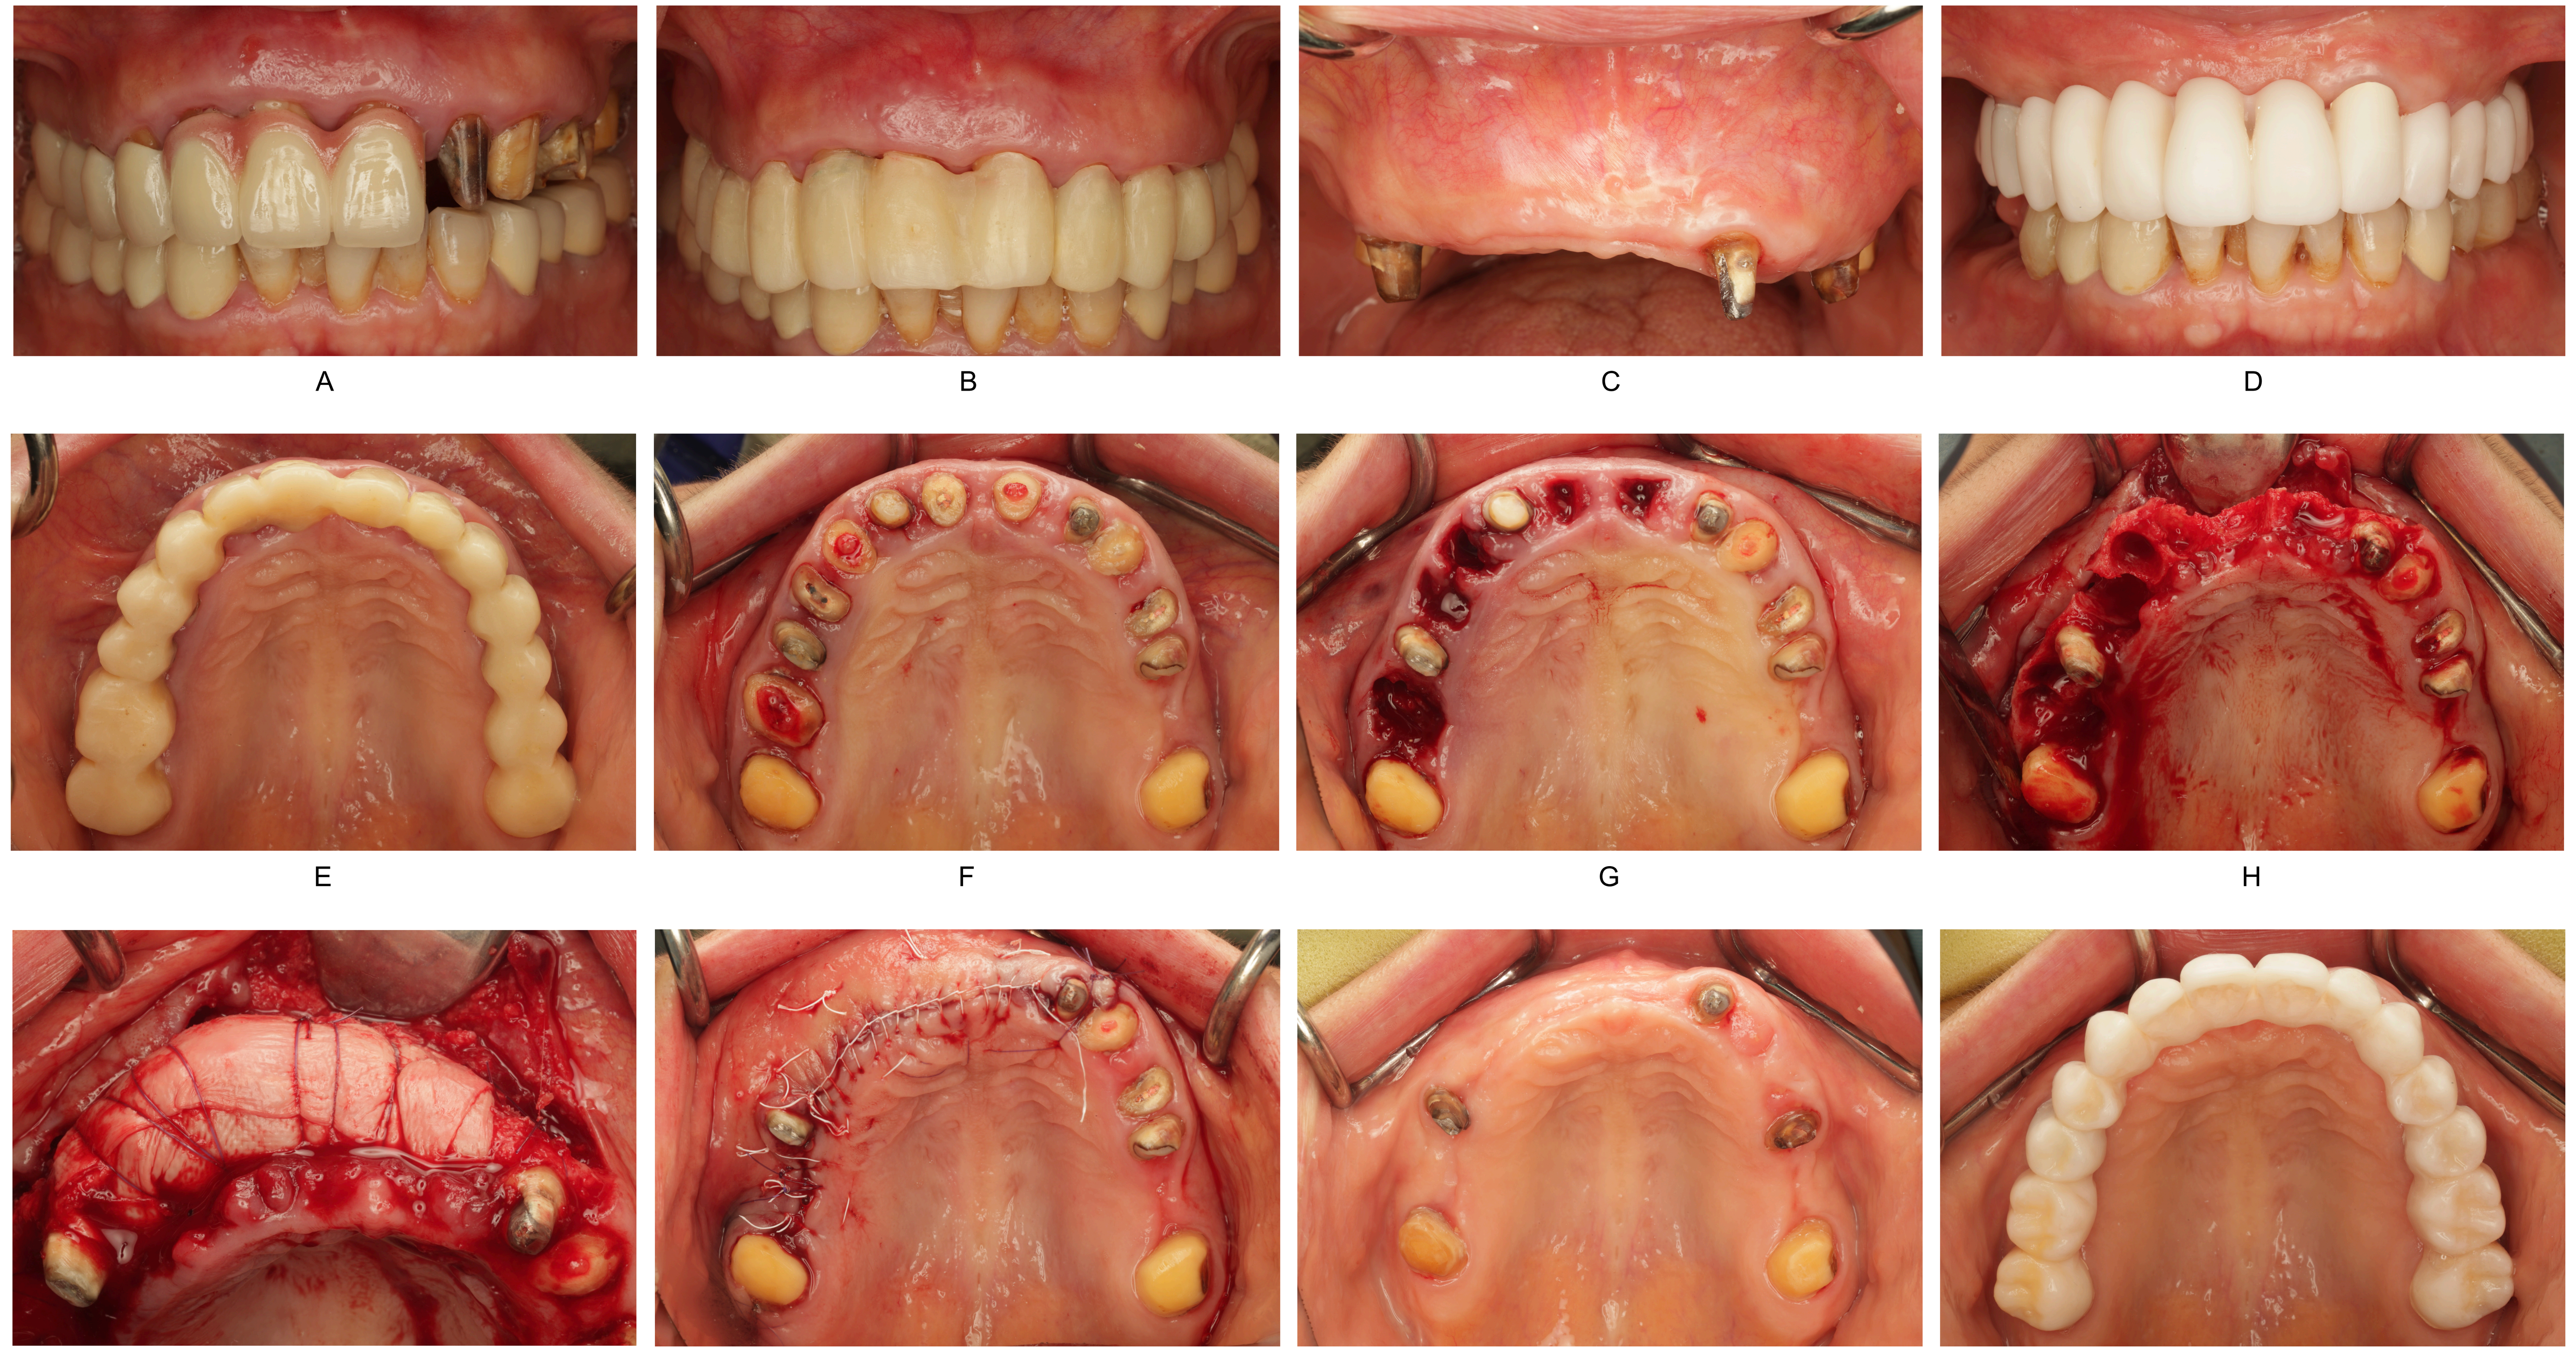
A
B
C
D
E
F
G
H
I
J
K
L

Figure 2. (A) Initial frontal view of failing prosthesis. (B) Long-span provisional delivered after removal of the previous fixed prosthesis. (C) 8-month postoperative frontal view of remaining abutment teeth supporting the provisional. (D) Frontal view 8 months after extractions and GBR, showing new provisional correcting incisal edge position and occlusal plane. (E) Occlusal view with original long-span provisional. (F) Preoperative occlusal view. (G) Extractions of teeth #3, #5, #6, #8, and #9. (H) Flap elevation showing severe buccal bone dehiscence. (I) Guided bone regeneration with suture stabilization. (J) Immediate postoperative occlusal view. (K) 8-month postoperative occlusal view of remaining abutment teeth. (L) 8-month postoperative occlusal view with new provisional.

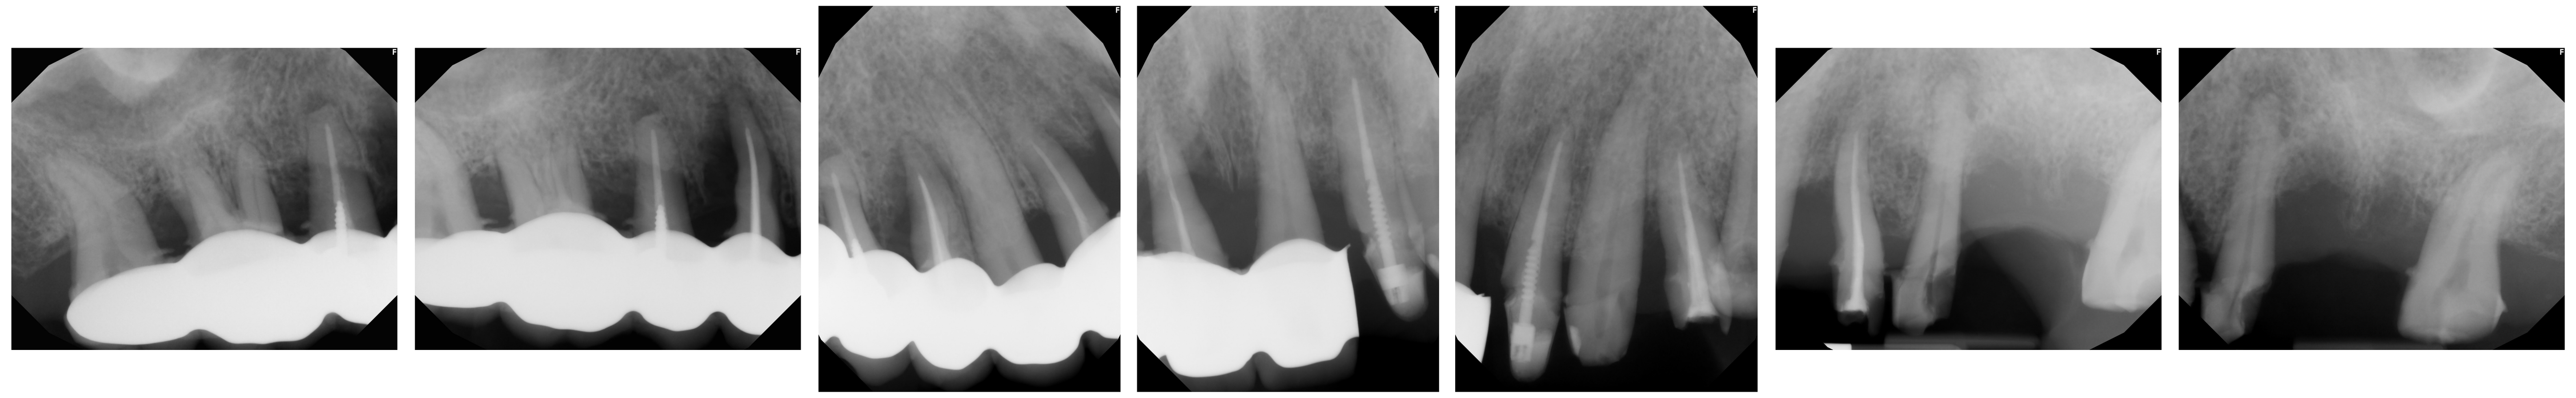
A
B
C
D
E
F
G

Figure 1. Initial maxillary radiographs indicate severe attachment loss, periapical pathosis, and deep carious lesion with pulp involvement.

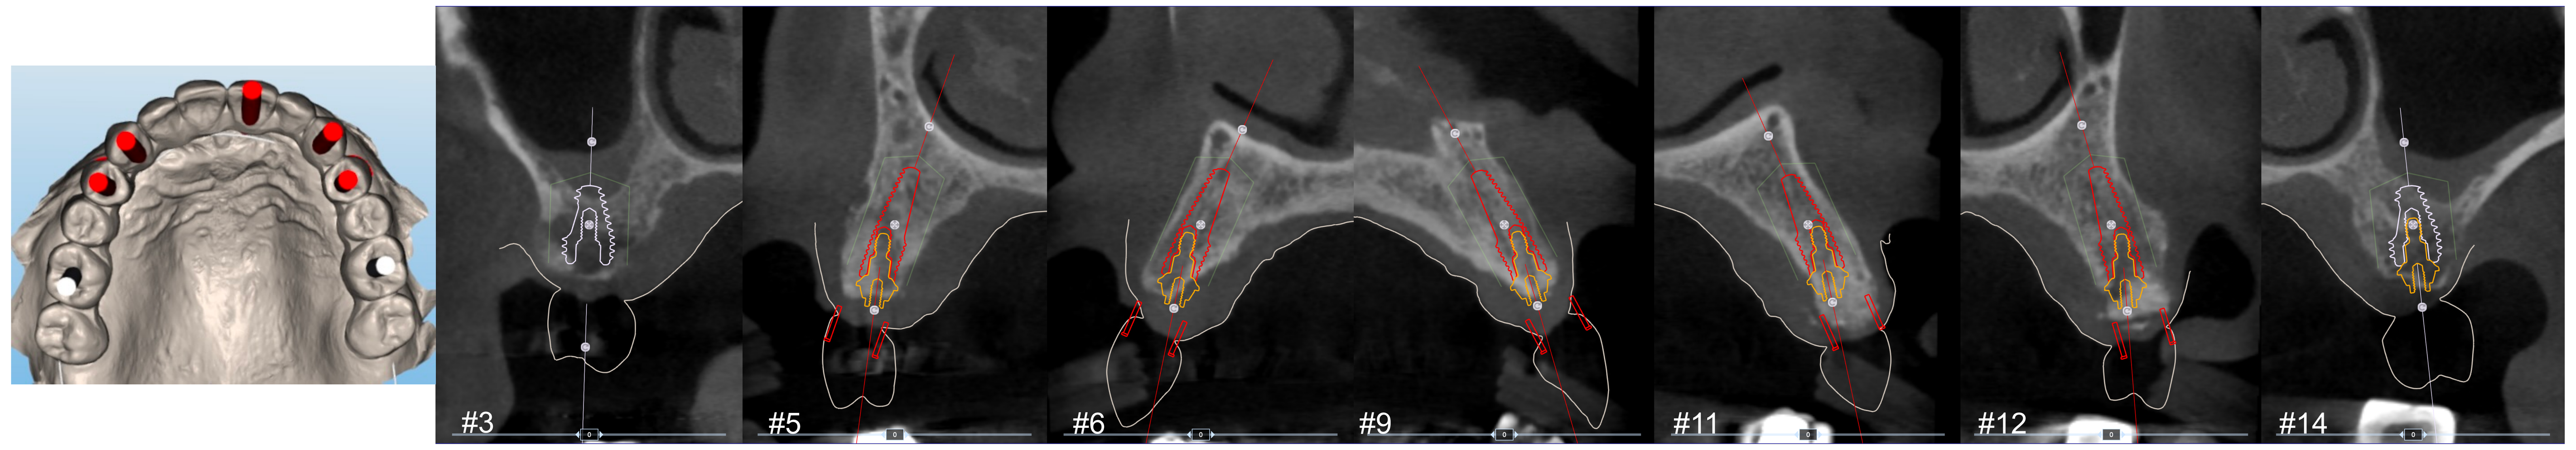
#3
#5
#6
#9
#11
#12
#14

Figure 3. Digital planning of implant placement using Southern Co-Axis and MAX implants to optimize angulation and achieve prosthetically driven positioning.

Reference

1. Mijiritsky E, Mazor Z, Lorean A, Mortellaro C, Mardinger O, Levin L. Transition from hopeless dentition to full-arch fixed-implant-supported rehabilitation by a staged extraction approach: rationale and technique. *J Craniofac Surg*. 2014;25(3):847-850.
2. Cordaro L, Torsello F, Ercoli C, Gallucci G. Transition from failing dentition to a fixed implant-supported restoration: a staged approach. *Int J Periodontics Restorative Dent*. 2007;27(5):481-487.

Penn Dental Medicine
University of Pennsylvania