Impact of ultrasonography,teleradiology,and free health insurance in resource-limited settings

Shambhu Dutta Joshi, Dinesh Joshi, Mahabaudha Medical Center, Dhangadhi, Nepal

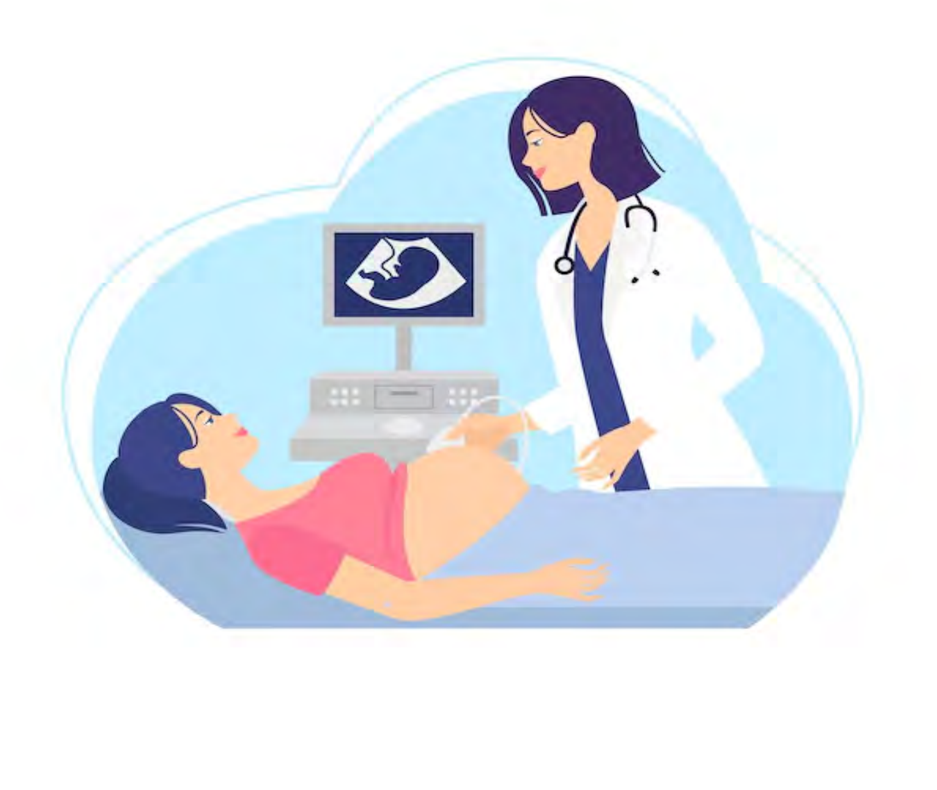

ISUOG WORLD CONGRESS
14–17 SEPTEMBER 2025
CANCUN, MEXICO

Objectives:

To assess the contribution of USG, facilitated by teleradiology and in detecting congenital anomalies and infections among pregnant women as a Point of Care USG.

Methods:

Retrospective study analysed on **300 pregnancies**.
Among these, **60 were high-risk pregnancies** where the mother had an **STI or was HIV positive**.
Data analysis performed using SPSS.

Results:

USG detected anomalies in **4.2%** pregnancies (17% neural tube defects)
Anomalies in high-risk STIs/HIV pregnancies: **6.8%**
Teleradiology → **85% rise** in referral rates in remote areas
Intrauterine infections: **9.1%** (congenital syphilis: 1.4%)
Among HIV+ mothers: USG **92.5% sensitivity** for placental abnormalities vs clinical exam
ANC adherence: **40%** → **68%** with free ANM-conducted USG
Free scans saved **USD 13 per patient**
Rural–urban diagnostic concordance: **94%**
Predictors of better outcomes:
Free USG scan (**OR=2.3**)
3D/4D teleradiology (**OR=1.8**)

Conclusions:

The integration of above services has been able to drastically improve the diagnosis and management of congenital anomalies and infections. This model is scalable and sustainable in resource-limited contexts by enhancing ANC adherence and improving diagnostic efficiency in the rural setting. More investment in the training of ANMs and expanding teleradiology networking and health-insurance schemes will be necessary to maintain such gains. Government and Local stakeholders should regular aware for USG gender identity disclosure which is illegal

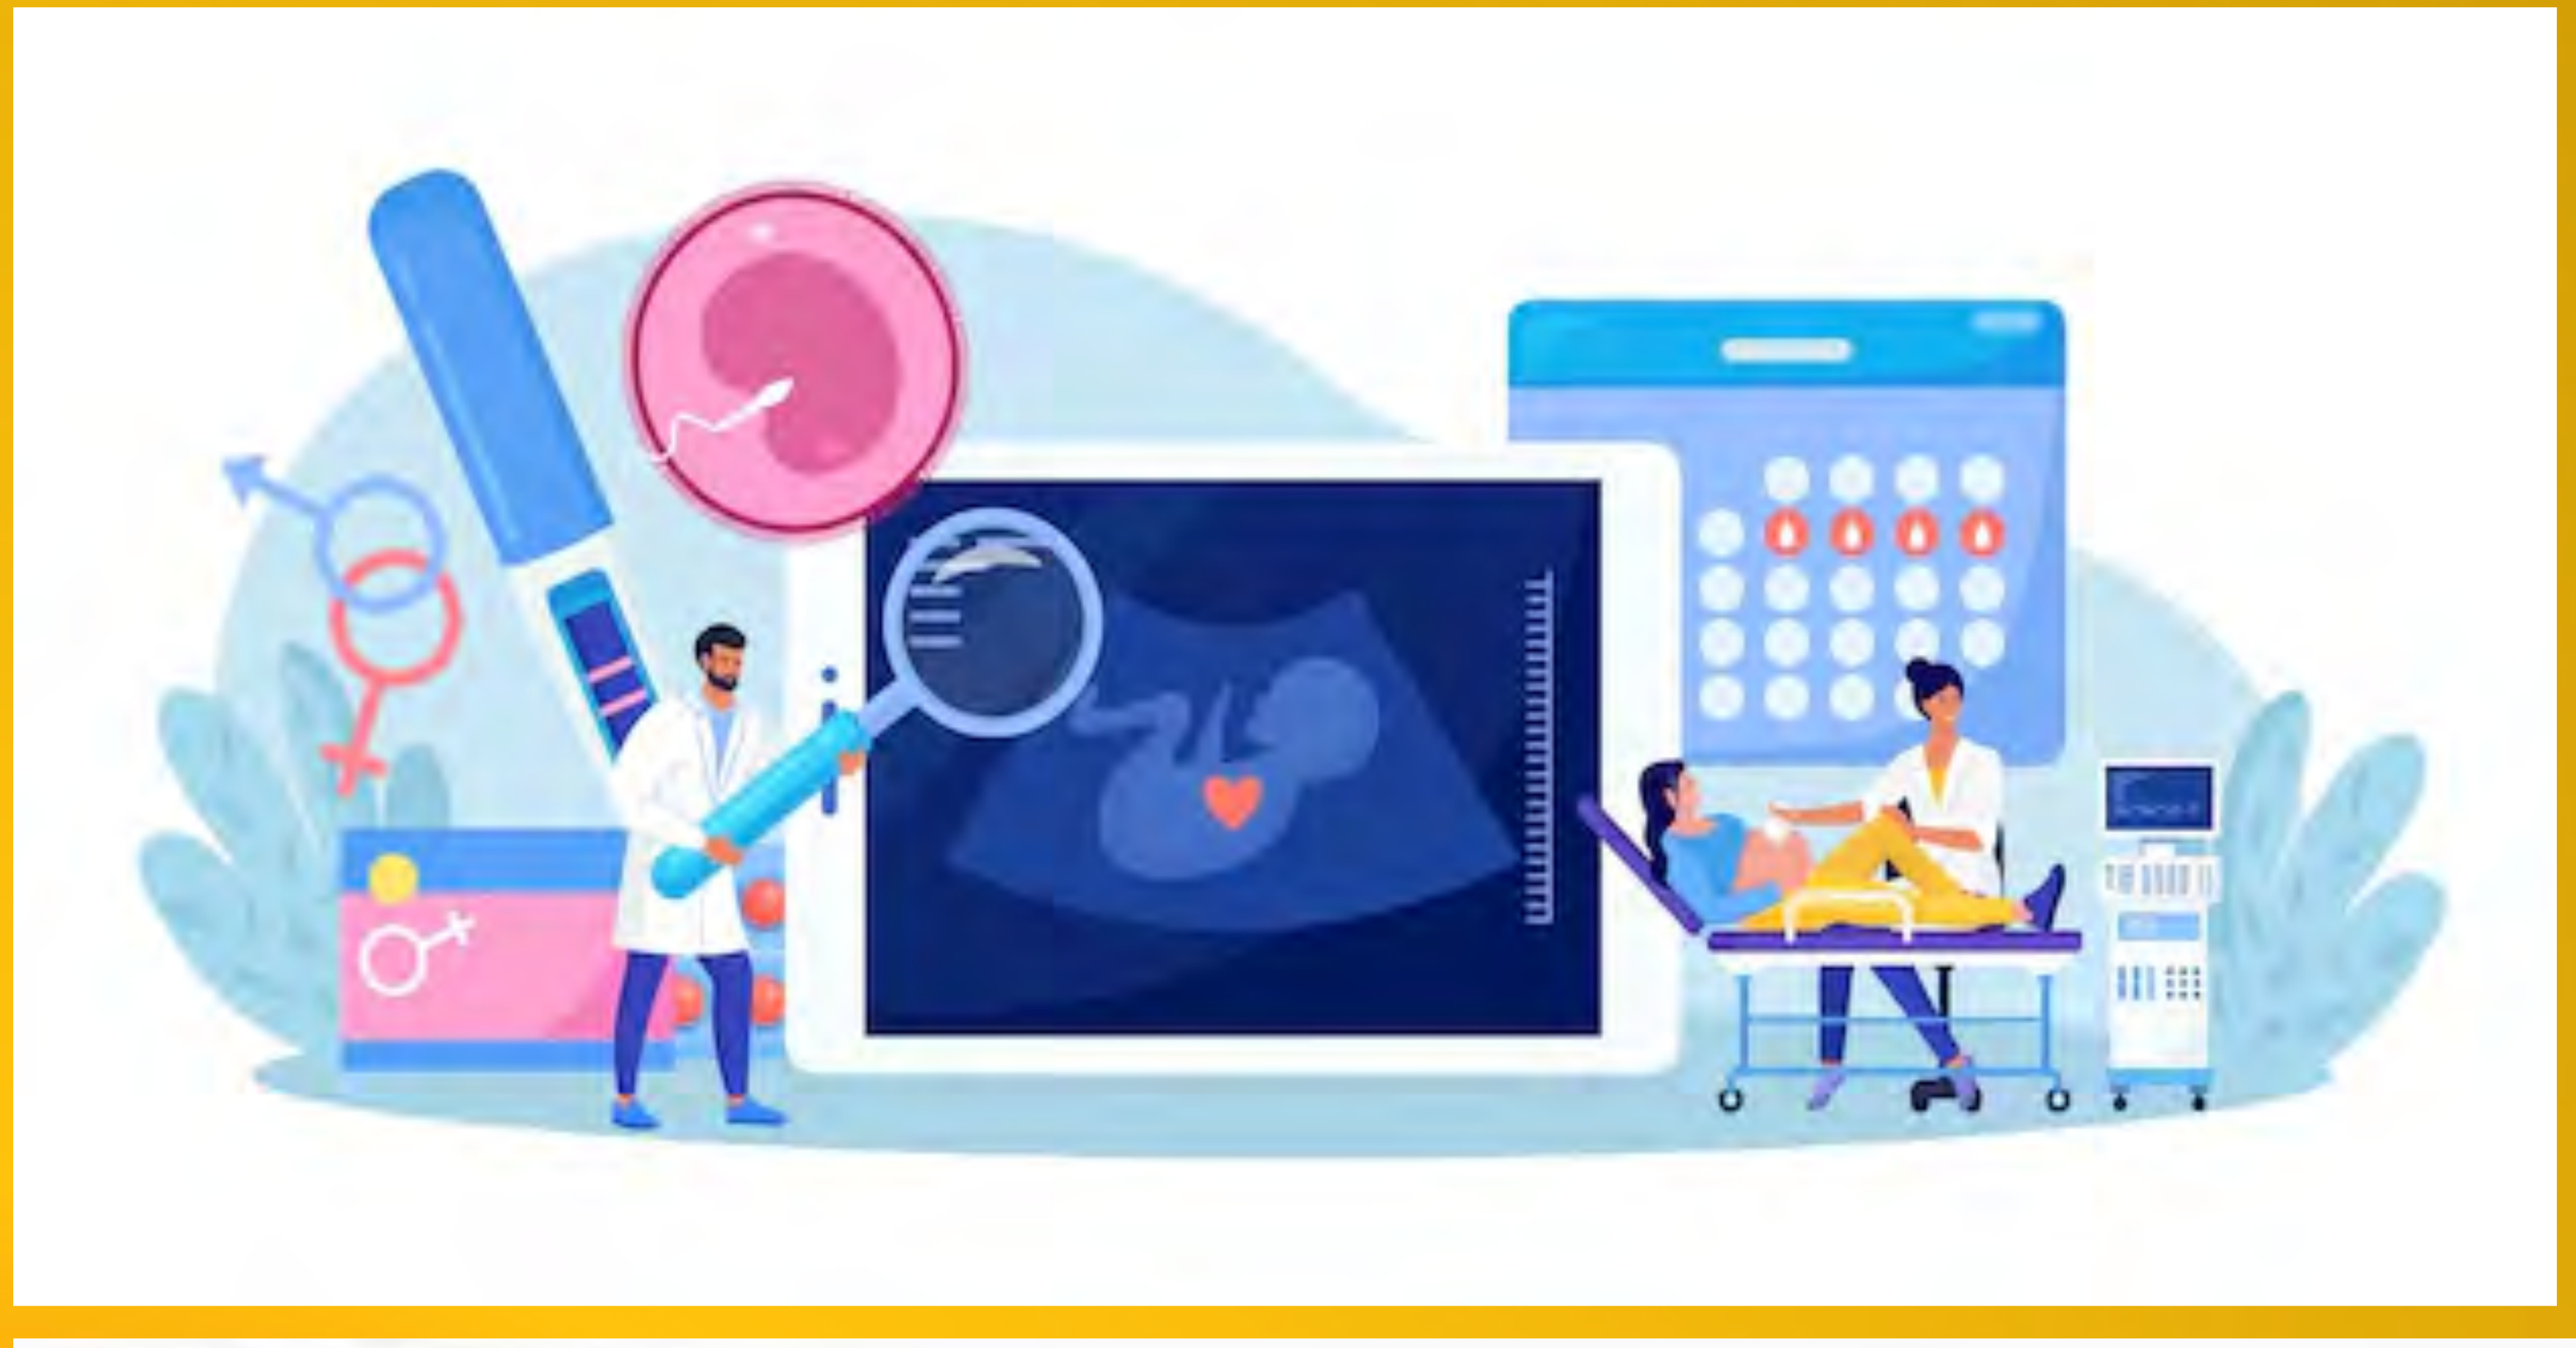

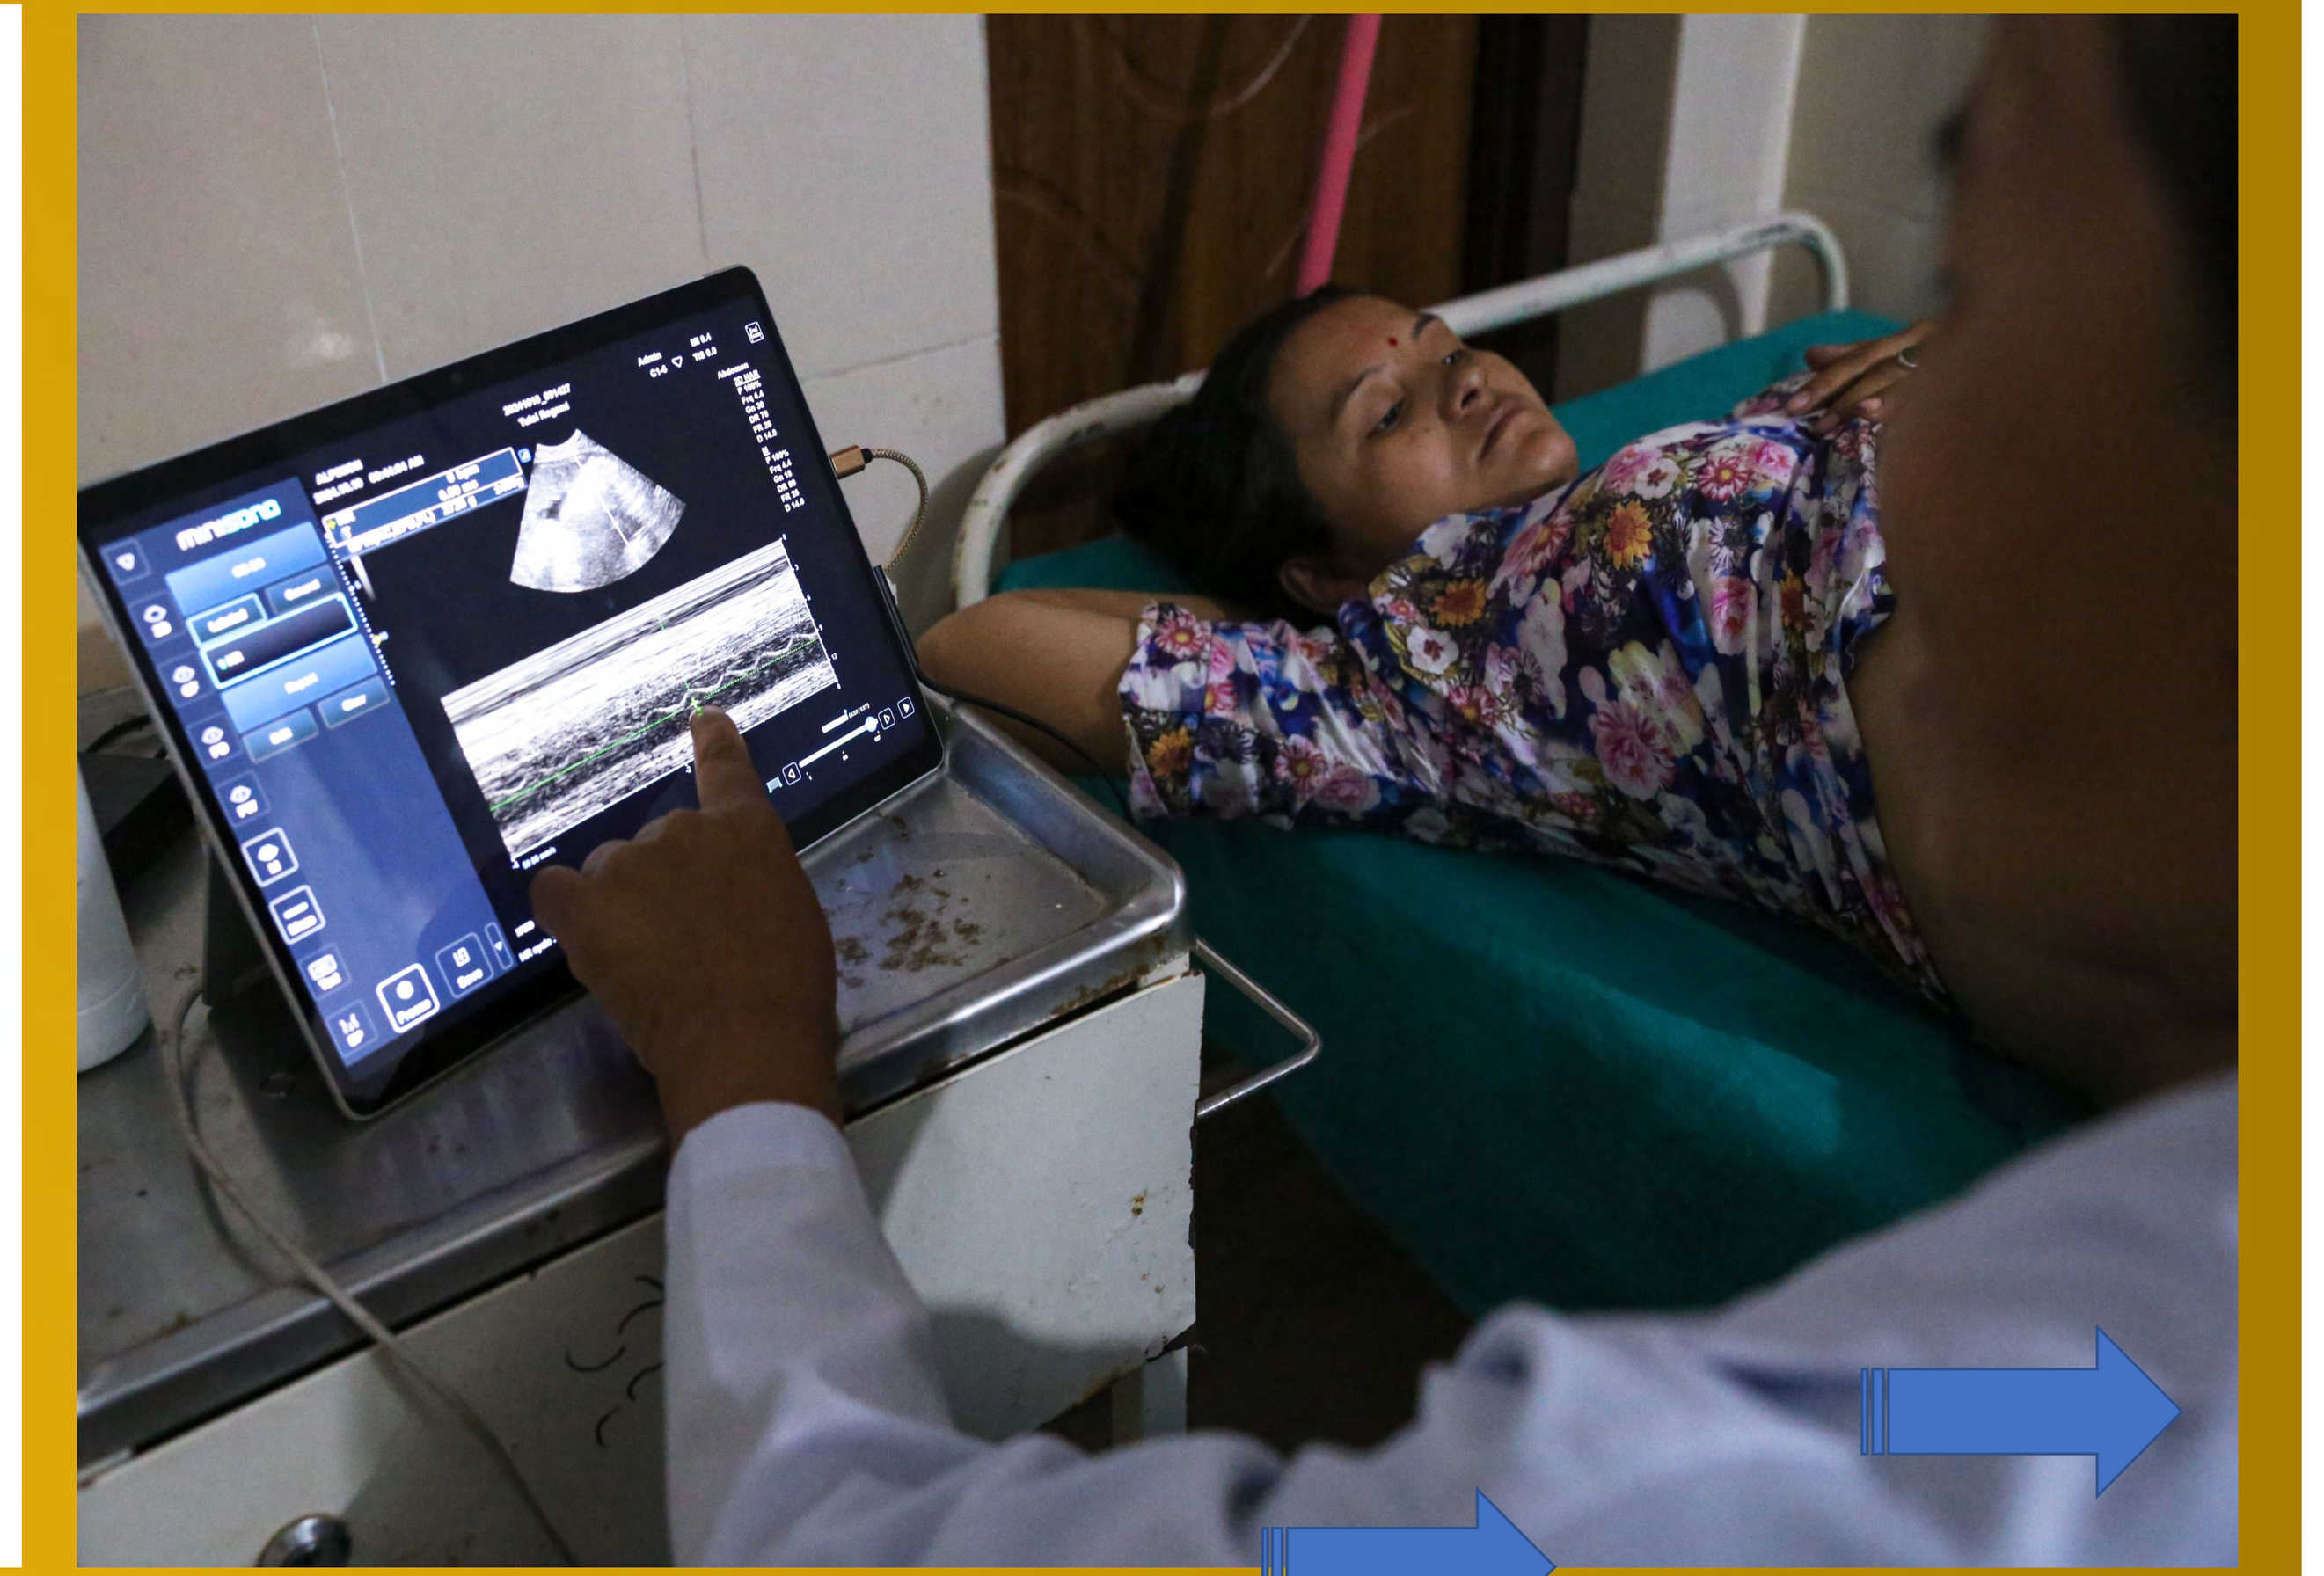

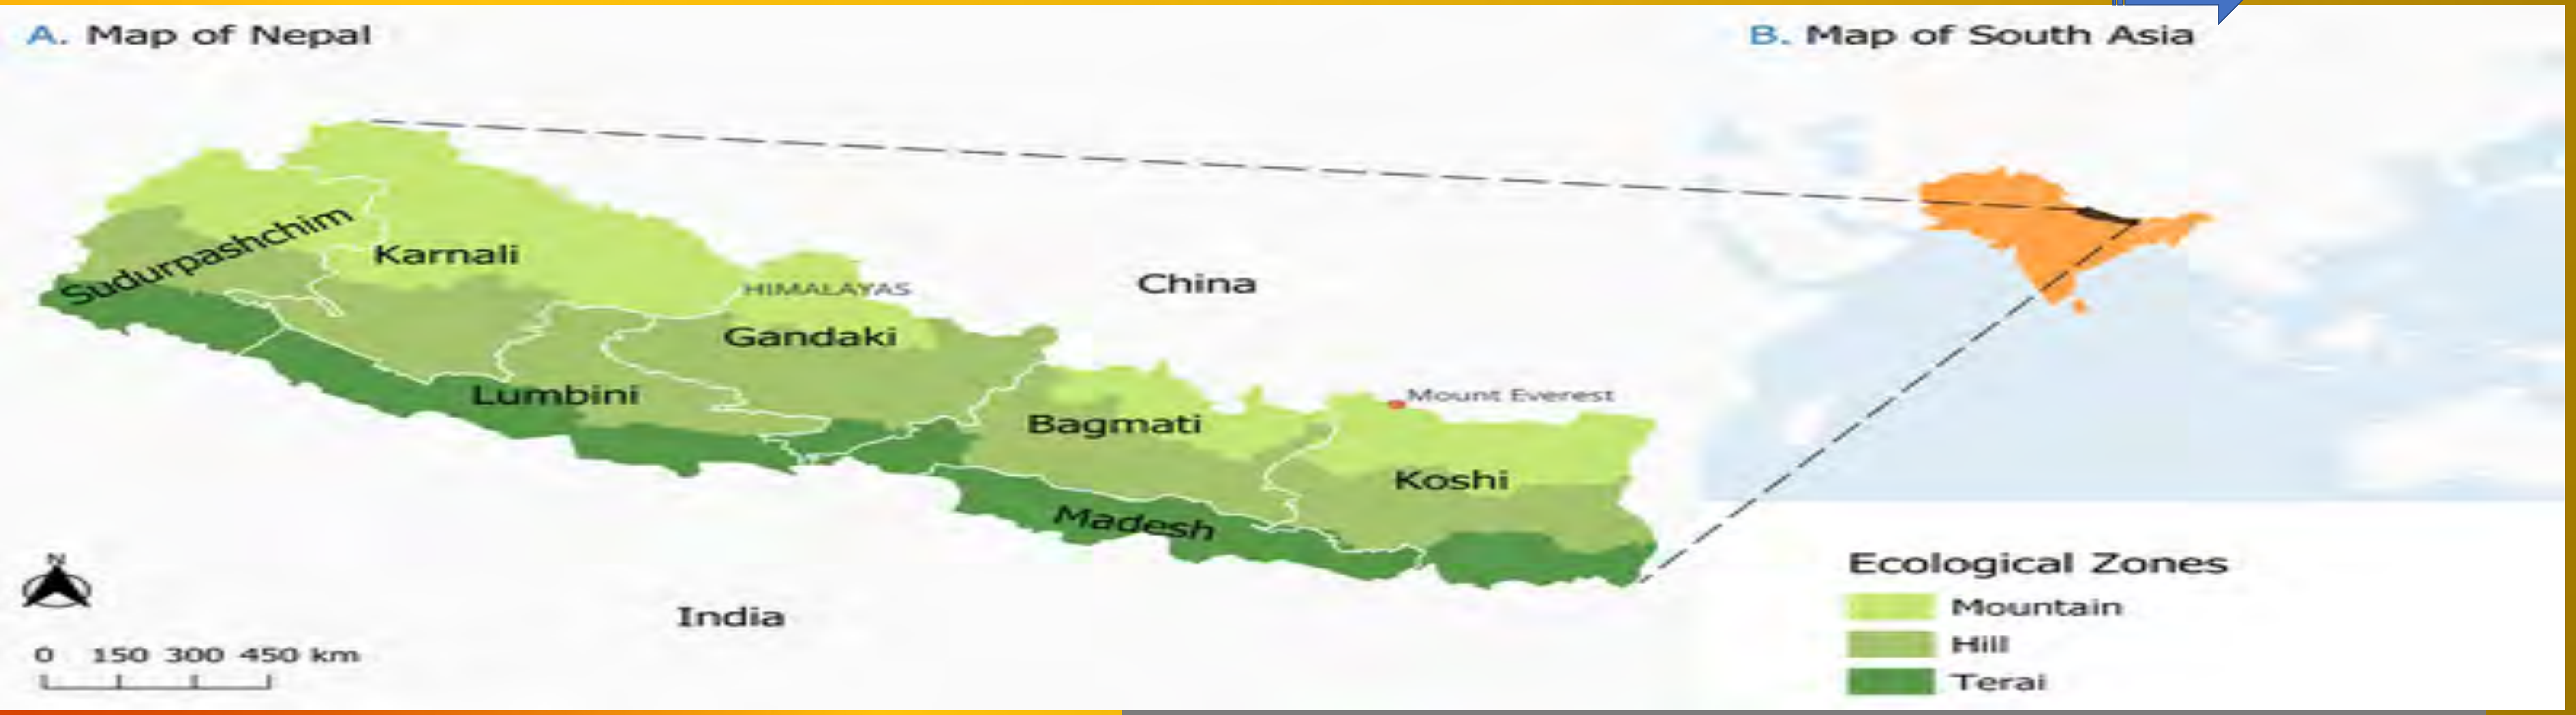
A. Map of Nepal
B. Map of South Asia
Sudurpashchim
Karnali
HIMALAYAS
Gandaki
China
Lumbini
Bagmati
Mount Everest
Koshi
Madesh
India
0 150 300 450 km
Ecological Zones
Mountain
Hill
Terai

Conflict of Interest : None