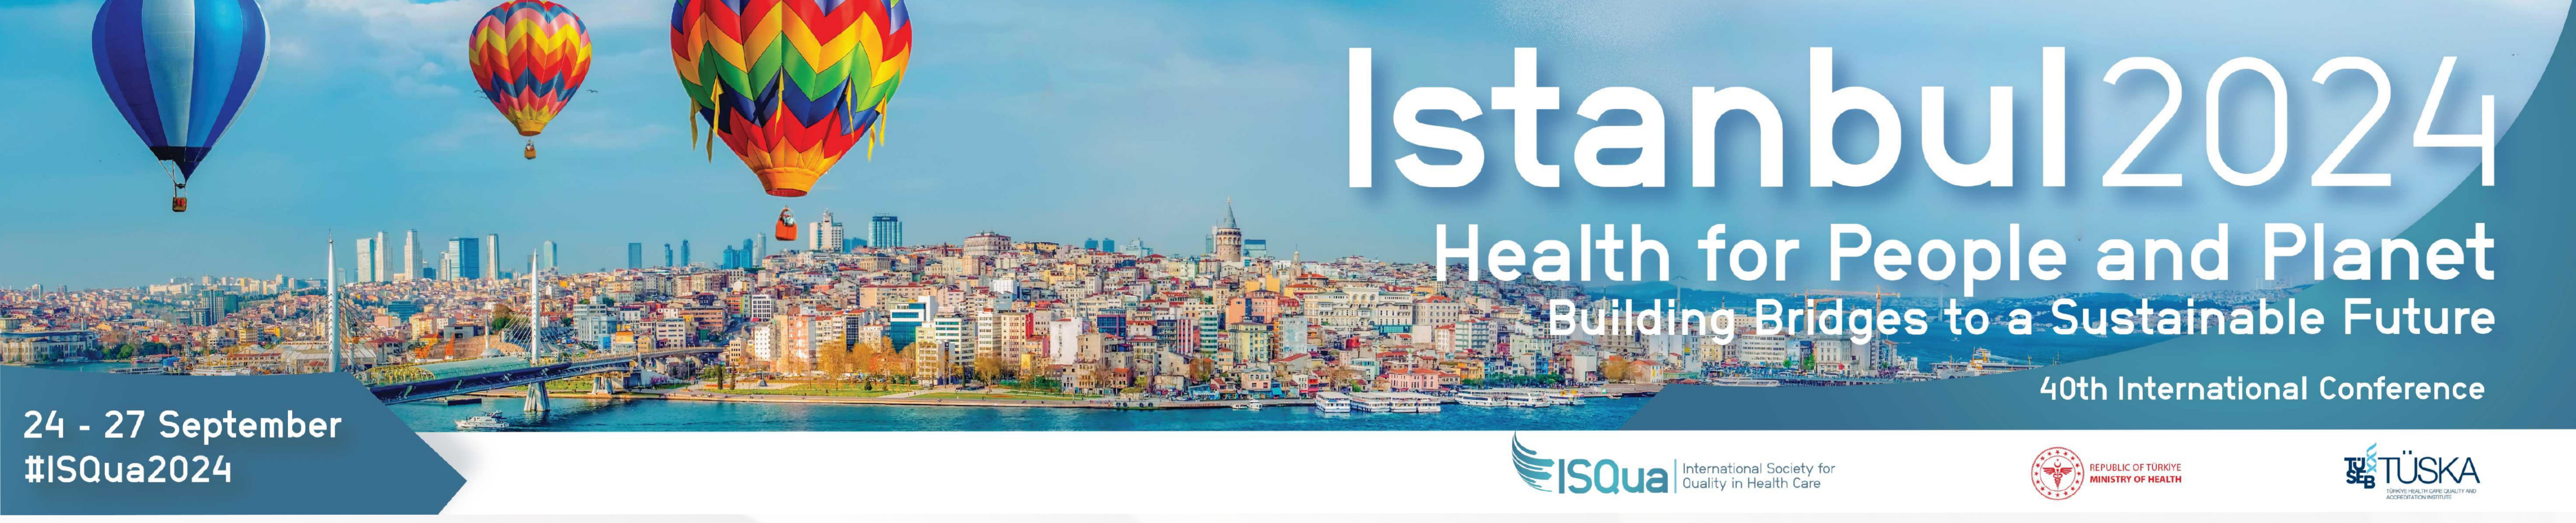
Istanbul2024
Health for People and Planet
Building Bridges to a Sustainable Future
40th International Conference
24 - 27 September
#ISQua2024
ISQua International Society for Quality in Health Care
REPUBLIC OF TÜRKIYE MINISTRY OF HEALTH
TÜSKA

A HOSPITAL LIKE NO OTHER, A CO PRODUCTION IN INDONESIA, WHEN CLINICAL CARE MEETS OUTSTANDING PATIENT AND FAMILY EXPERIENCE, A PLANETREE QUALITY IMPROVEMENT MODEL IN MANDAYA ROYAL HOSPITAL PURI.

Anastina Tahjoo*[1], Jeffrey Braithwaite[2], Hanny Rono[3], Sonny A. Wijaya[1] , Billiyam[4] , Khairul[4]
[1]Member, ISQua, Jakarta, Indonesia, [2]Past President, ISQua, Sydney, Australia, [3]Board Member, ISQua & KARS, [4]Marketing Communication, Mandaya Royal Hospital Puri, Jakarta, Indonesia

P-249

INTRODUCTION

DISCOVER

Indonesia has made significant progress in healthcare services over the past few decades. However, there is still a demand for international standard healthcare services in Indonesia which is often evident from the large number of patients traveling abroad for medical treatment. Mandaya, as a new hospital operating since July 2021, is designed to meet patients' growing needs and demands for high quality care.

However, capacity still lags behind demand.

Hospital Beds
- 1.18 : 1000 people
- Asia 3.3 : 1000 people
- OECD Countries 4.8 : 1000 people

Physicians
- 0.465 : 1000 people
- 2.294 : 1000 people
- OECD Countries 3.794 : 1000 people

Demand for Healthcare in Indonesia Expected to Rise

2010 x 240 million
+ 28%
2035 x 300 million

AIM

DEVELOP

To provide advanced medical care tailored to the individual needs and priorities of each patient with a focus not only on patient satisfaction, but more importantly, on the patient and family experience. Achieve the vision of patient and family experience by implementing the Planetree quality improvement model.

METHOD

GROW THRIVE

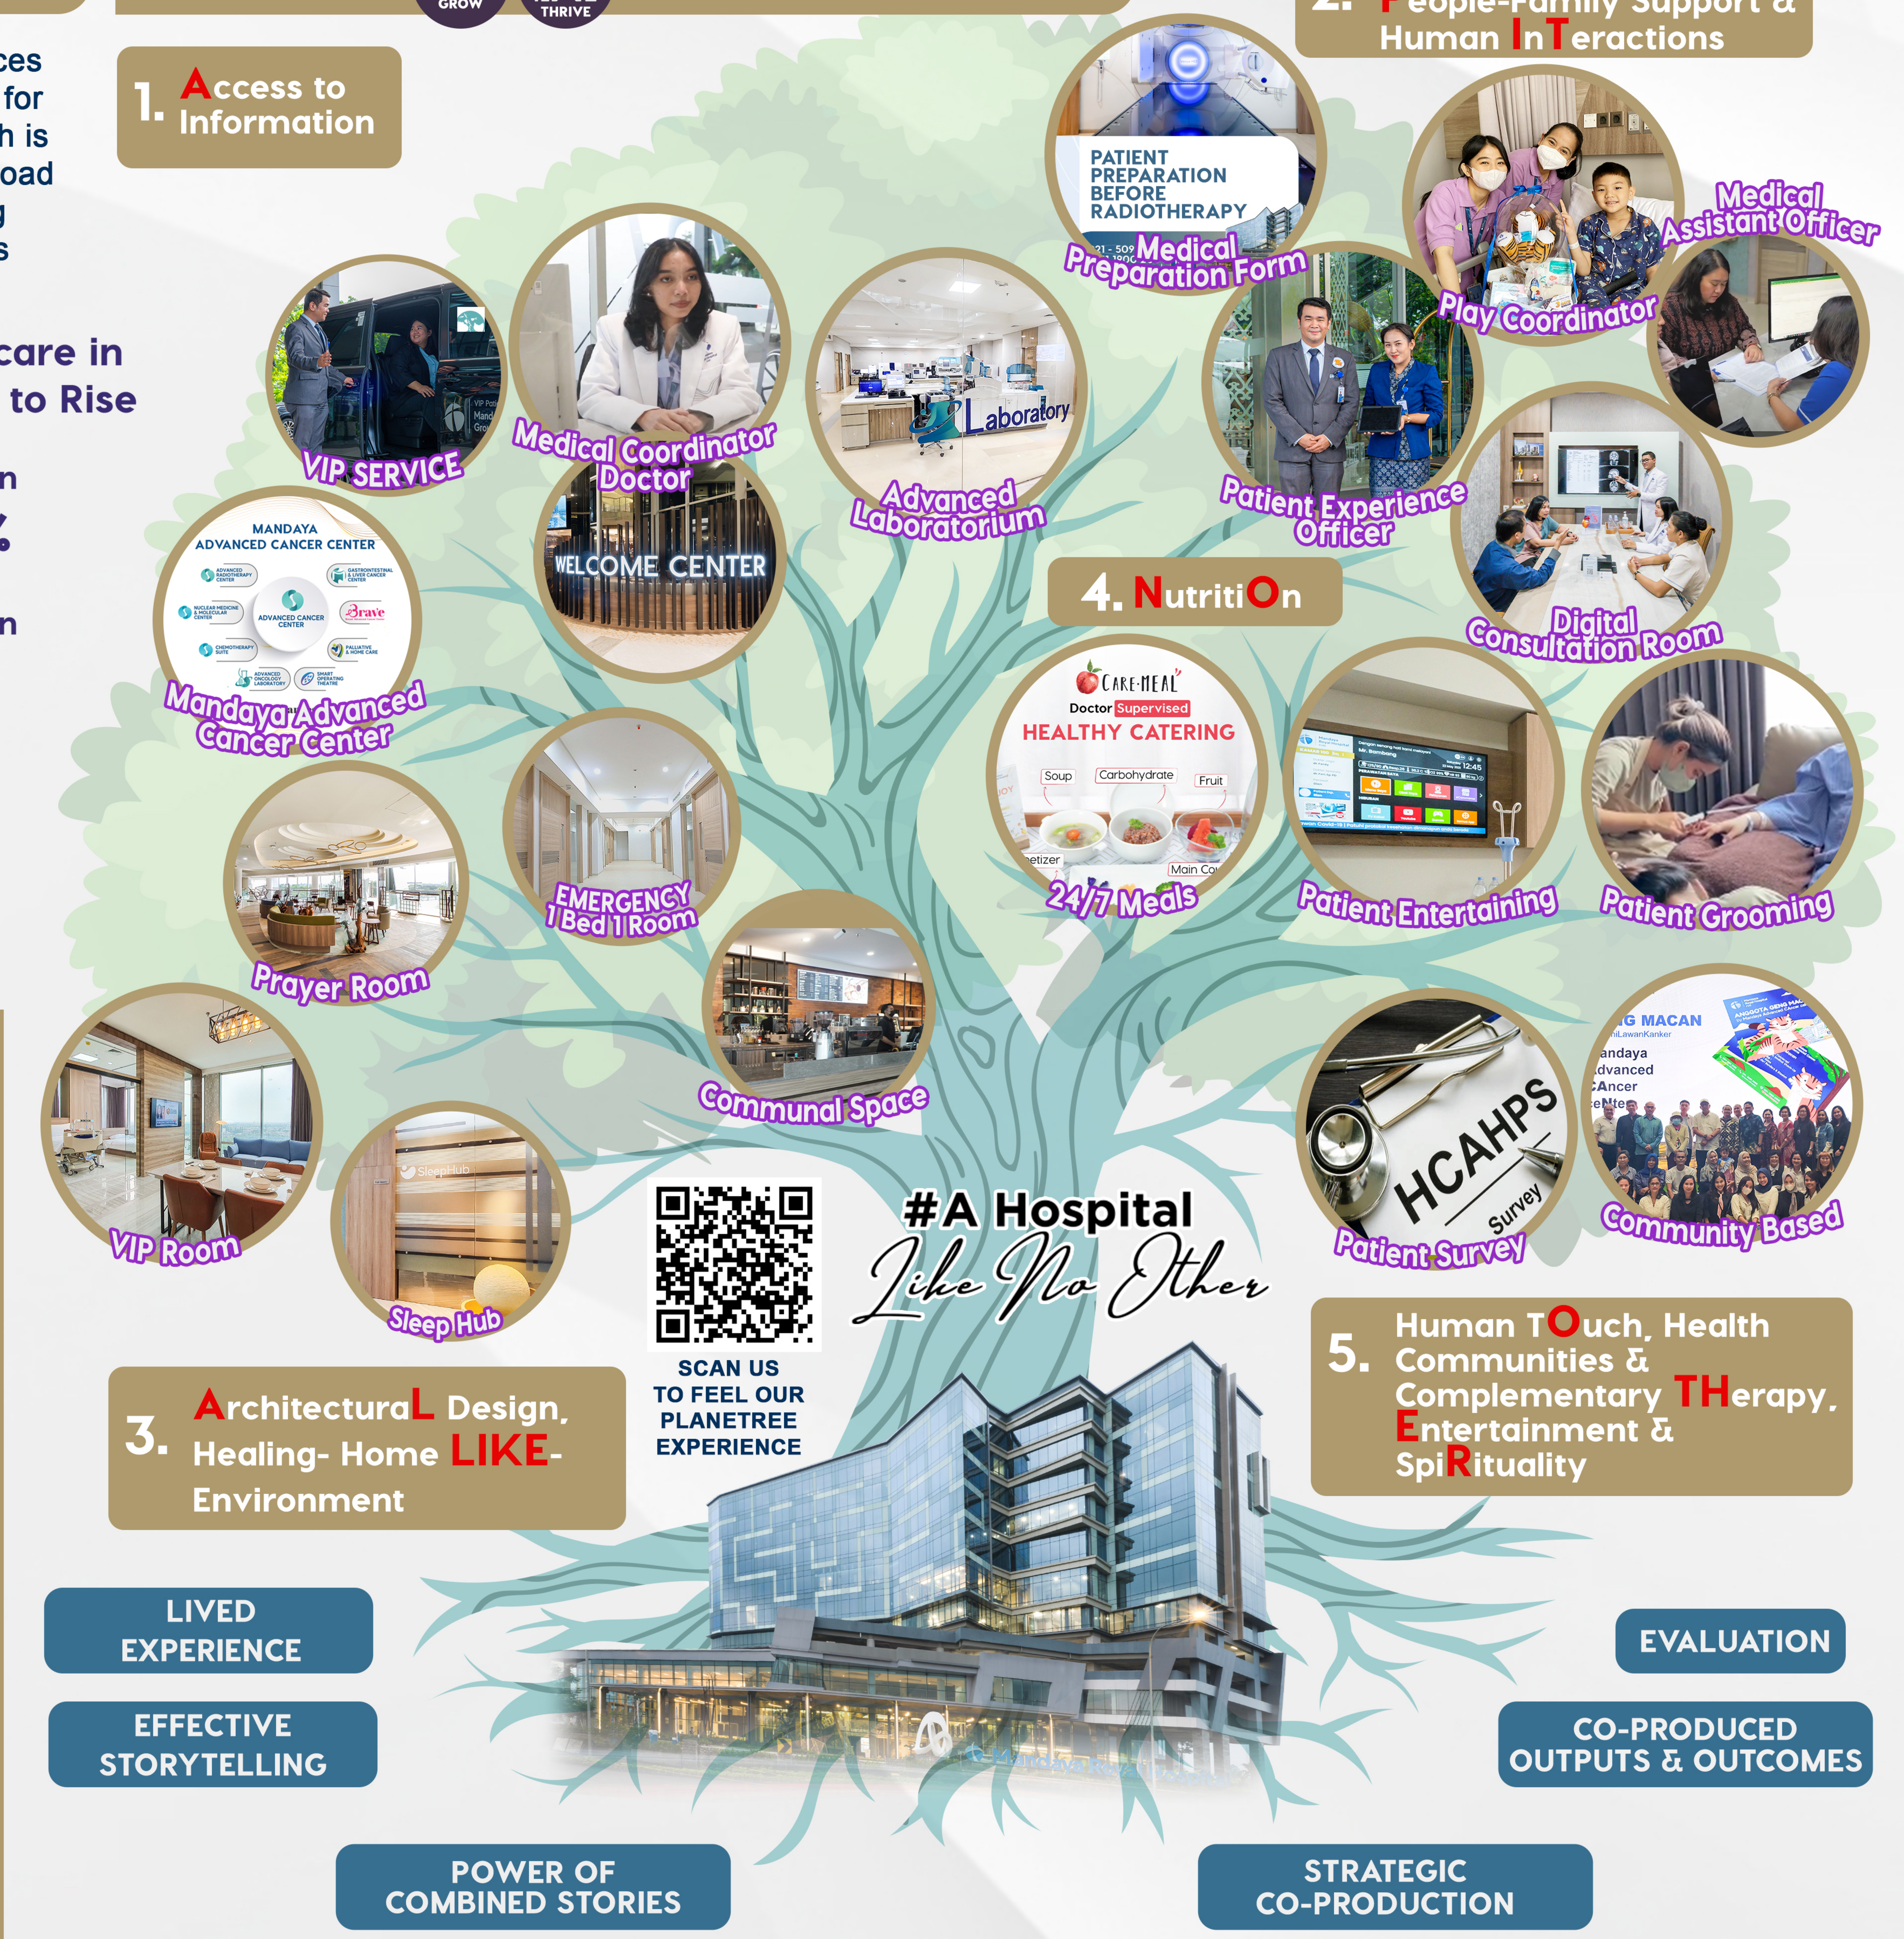
1. Access to Information
2. Human InteractiOnS, People-Family Support & Human InTeractions
PATIENT PREPARATION BEFORE RADIOTHERAPY
Medical Preparation Form
Play Coordinator
Medical Assistant Officer
VIP SERVICE
Medical Coordinator Doctor
Advanced Laboratorium
Patient Experience Officer
WELCOME CENTER
Digital Consultation Room
4. NutritiOn
CARE·MEAL Doctor Supervised HEALTHY CATERING
Soup
Carbohydrate
Fruit
24/7 Meals
Patient Entertaining
Patient Grooming
MANDAYA ADVANCED CANCER CENTER
Mandaya Advanced Cancer Center
EMERGENCY 1 Bed 1 Room
Prayer Room
Communal Space
VIP Room
Sleep Hub
HCAHPS Survey
Patient Survey
Community Based
SCAN US TO FEEL OUR PLANETREE EXPERIENCE
#A Hospital Like No Other
3. ArchitecturaL Design, Healing- Home LIKE- Environment
5. Human TOuch, Health Communities & Complementary THerapy, Entertainment & SpiRituality
LIVED EXPERIENCE
EFFECTIVE STORYTELLING
POWER OF COMBINED STORIES
STRATEGIC CO-PRODUCTION
EVALUATION
CO-PRODUCED OUTPUTS & OUTCOMES

RESULT

VALIDATE

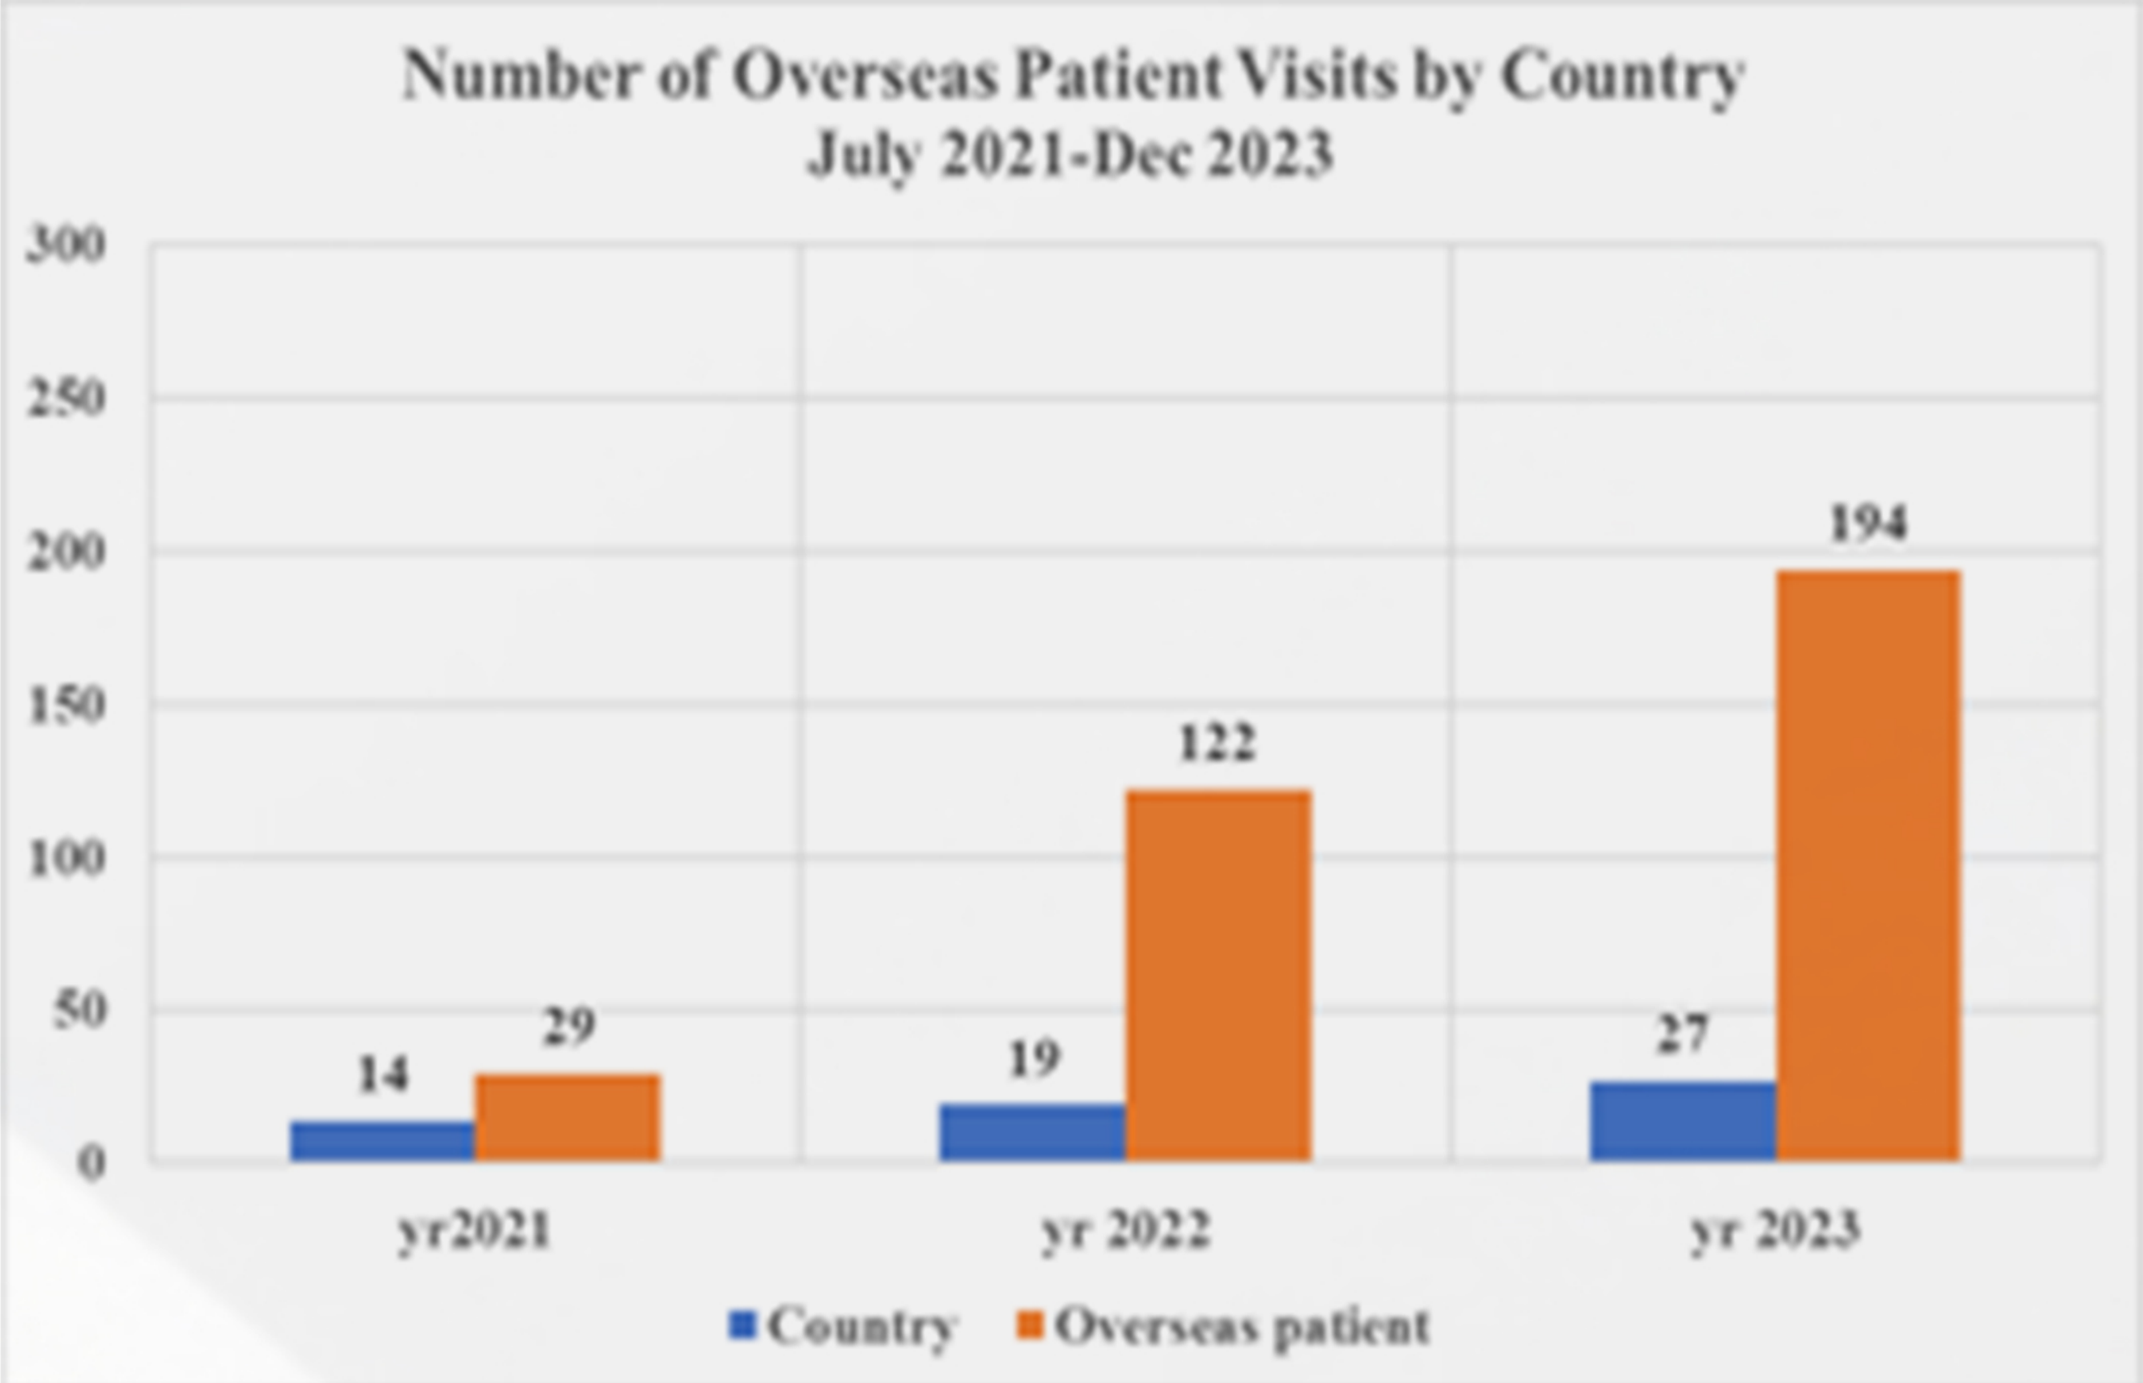
Number of Overseas Patient Visits by Country
July 2021-Dec 2023
yr2021: 14, 29
yr 2022: 19, 122
yr 2023: 27, 194
Country
Overseas patient

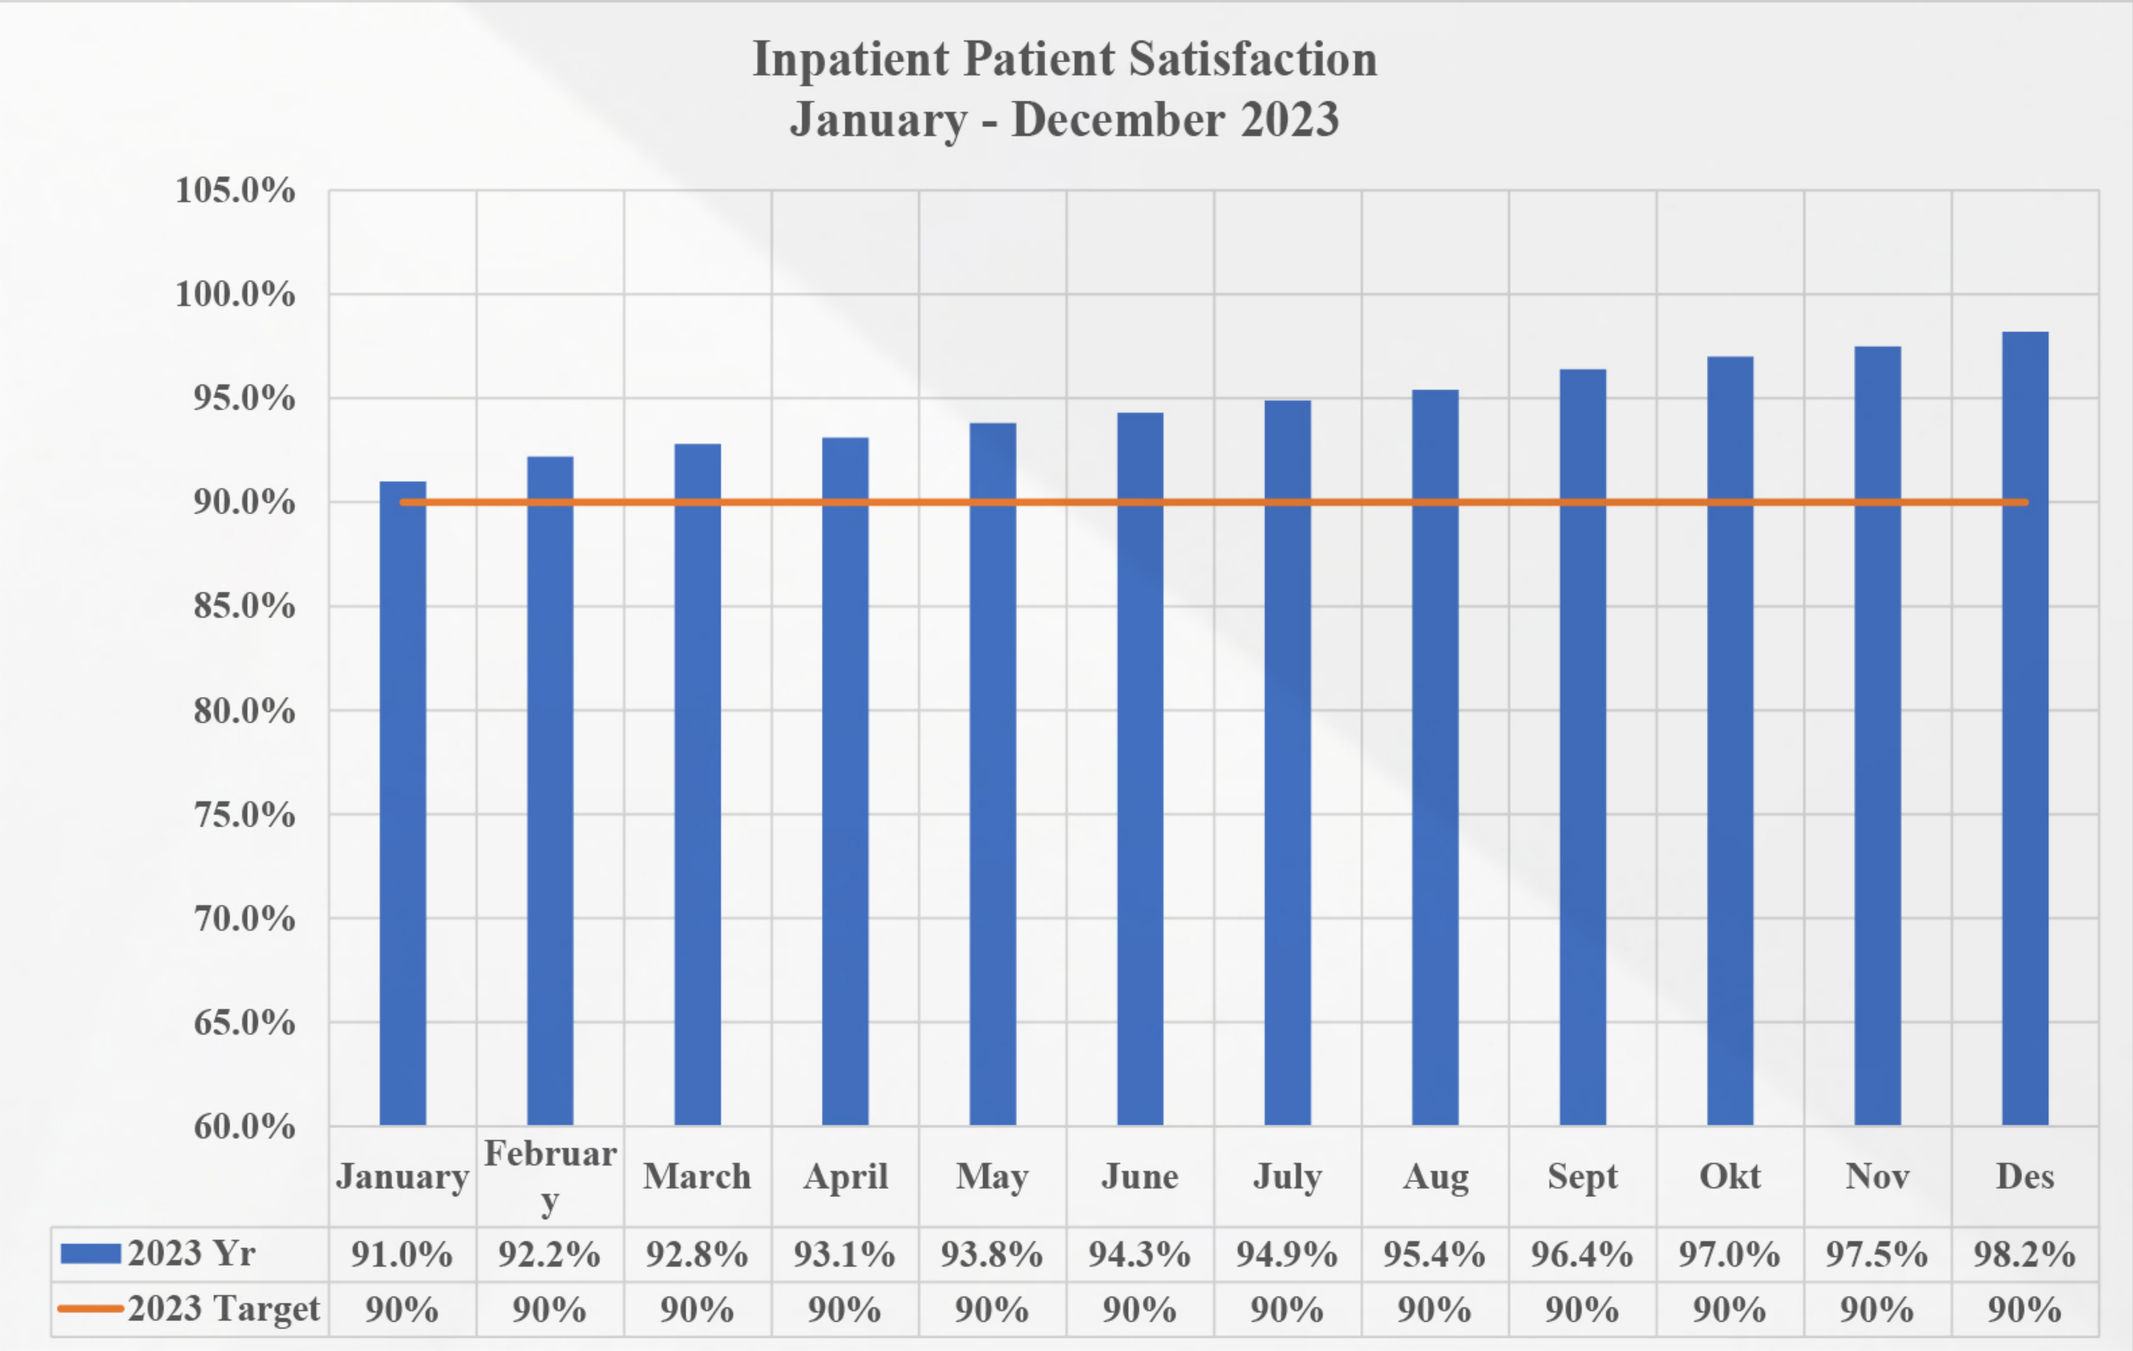
Inpatient Patient Satisfaction
January - December 2023

	January	February	March	April	May	June	July	Aug	Sept	Okt	Nov	Des
2023 Yr	91.0%	92.2%	92.8%	93.1%	93.8%	94.3%	94.9%	95.4%	96.4%	97.0%	97.5%	98.2%
2023 Target	90%	90%	90%	90%	90%	90%	90%	90%	90%	90%	90%	90%

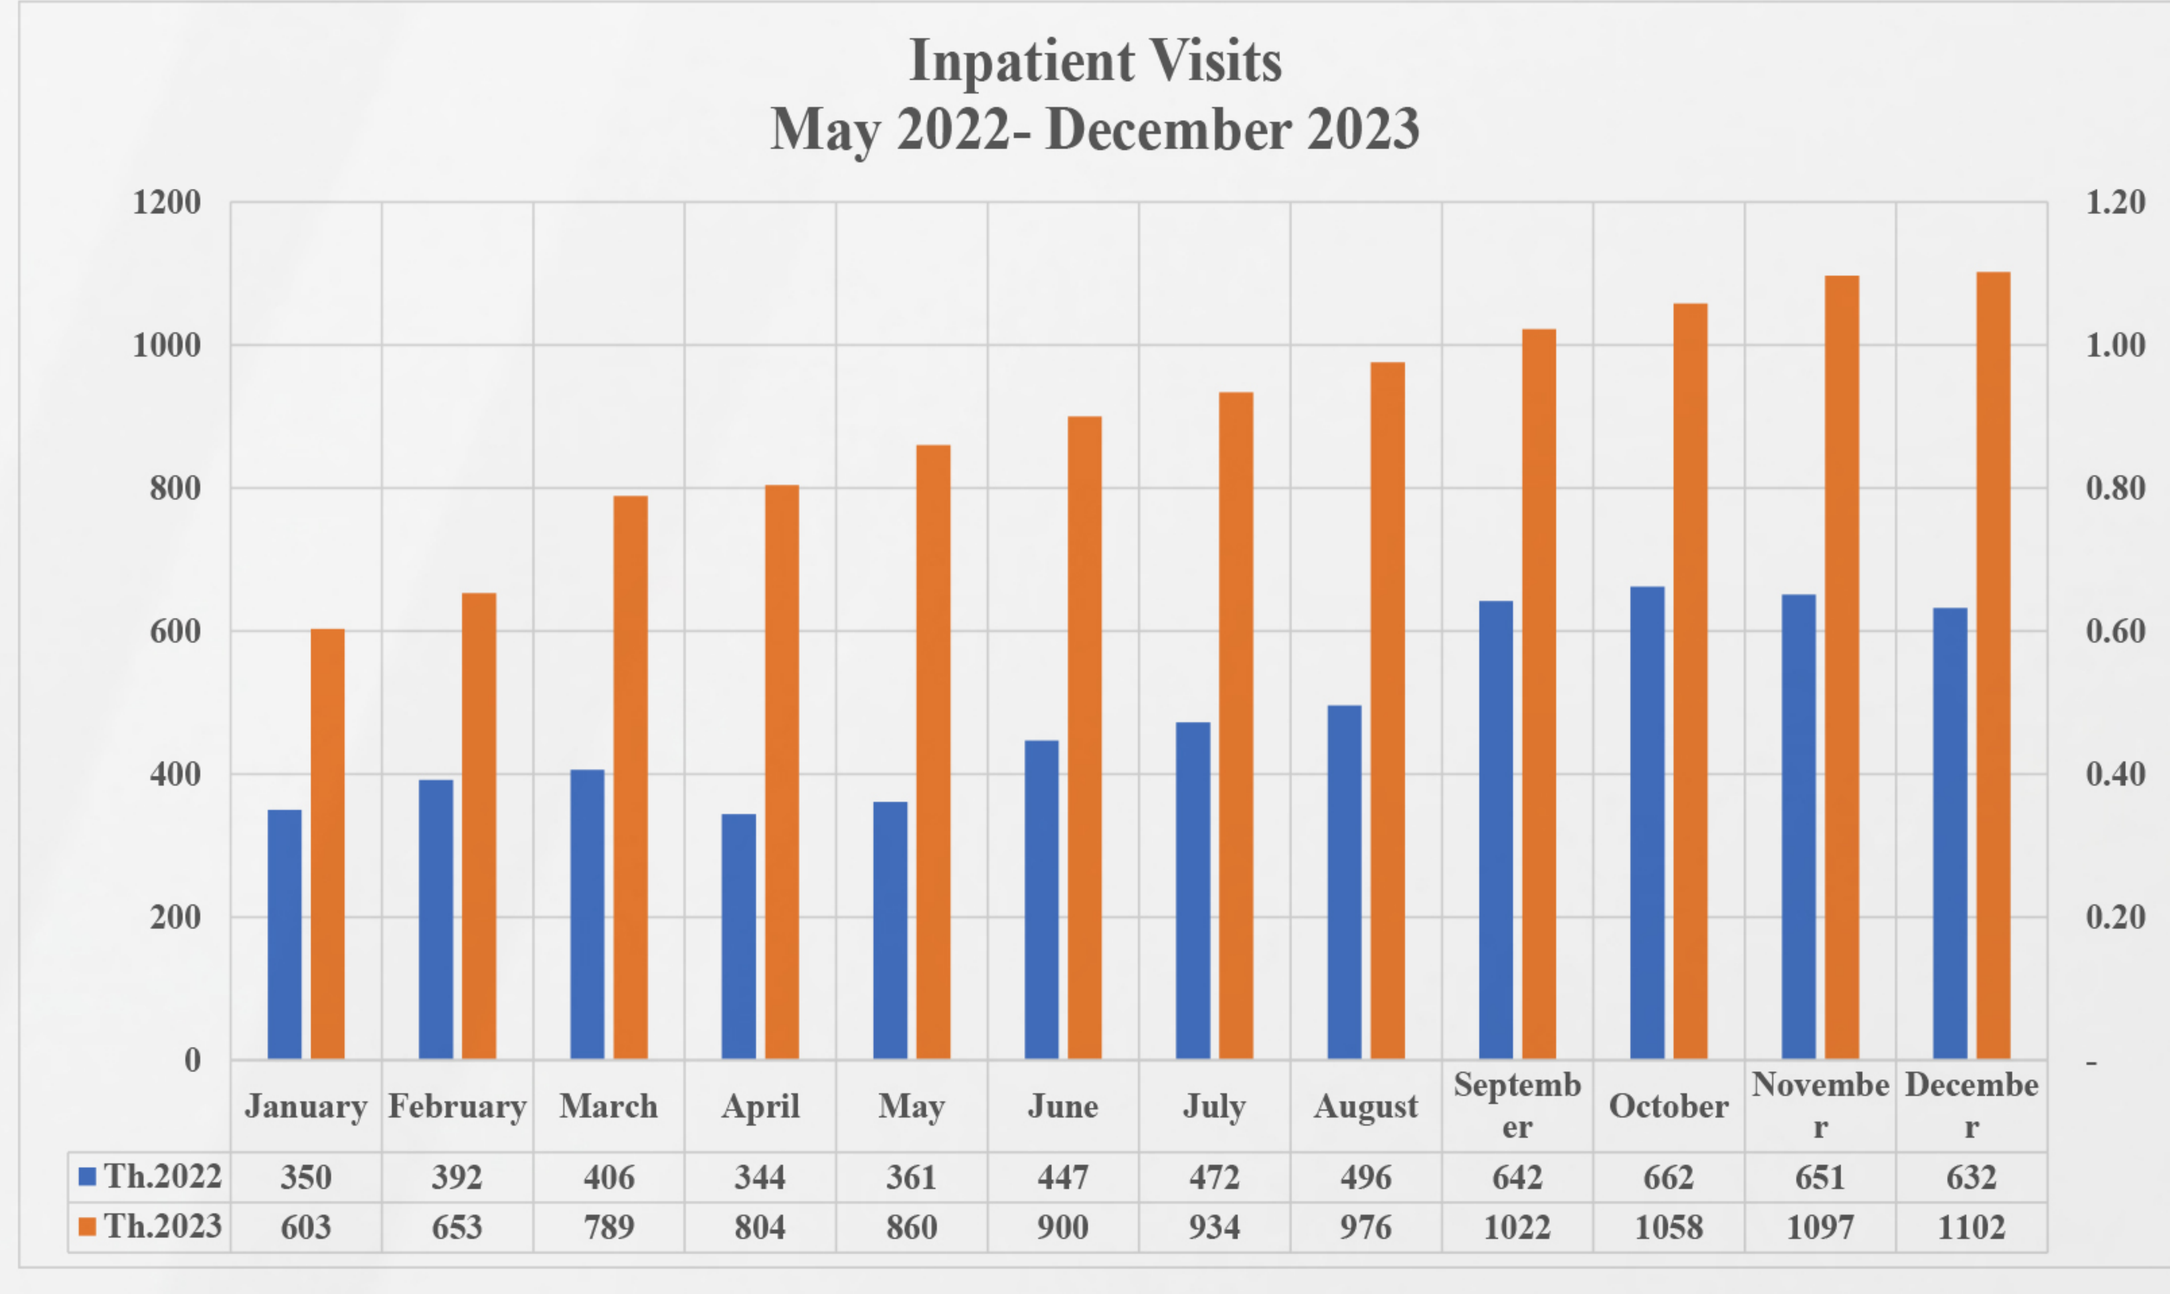
Inpatient Visits
May 2022- December 2023

	January	February	March	April	May	June	July	August	September	October	November	December
Th.2022	350	392	406	344	361	447	472	496	642	662	651	632
Th.2023	603	653	789	804	860	900	934	976	1022	1058	1097	1102

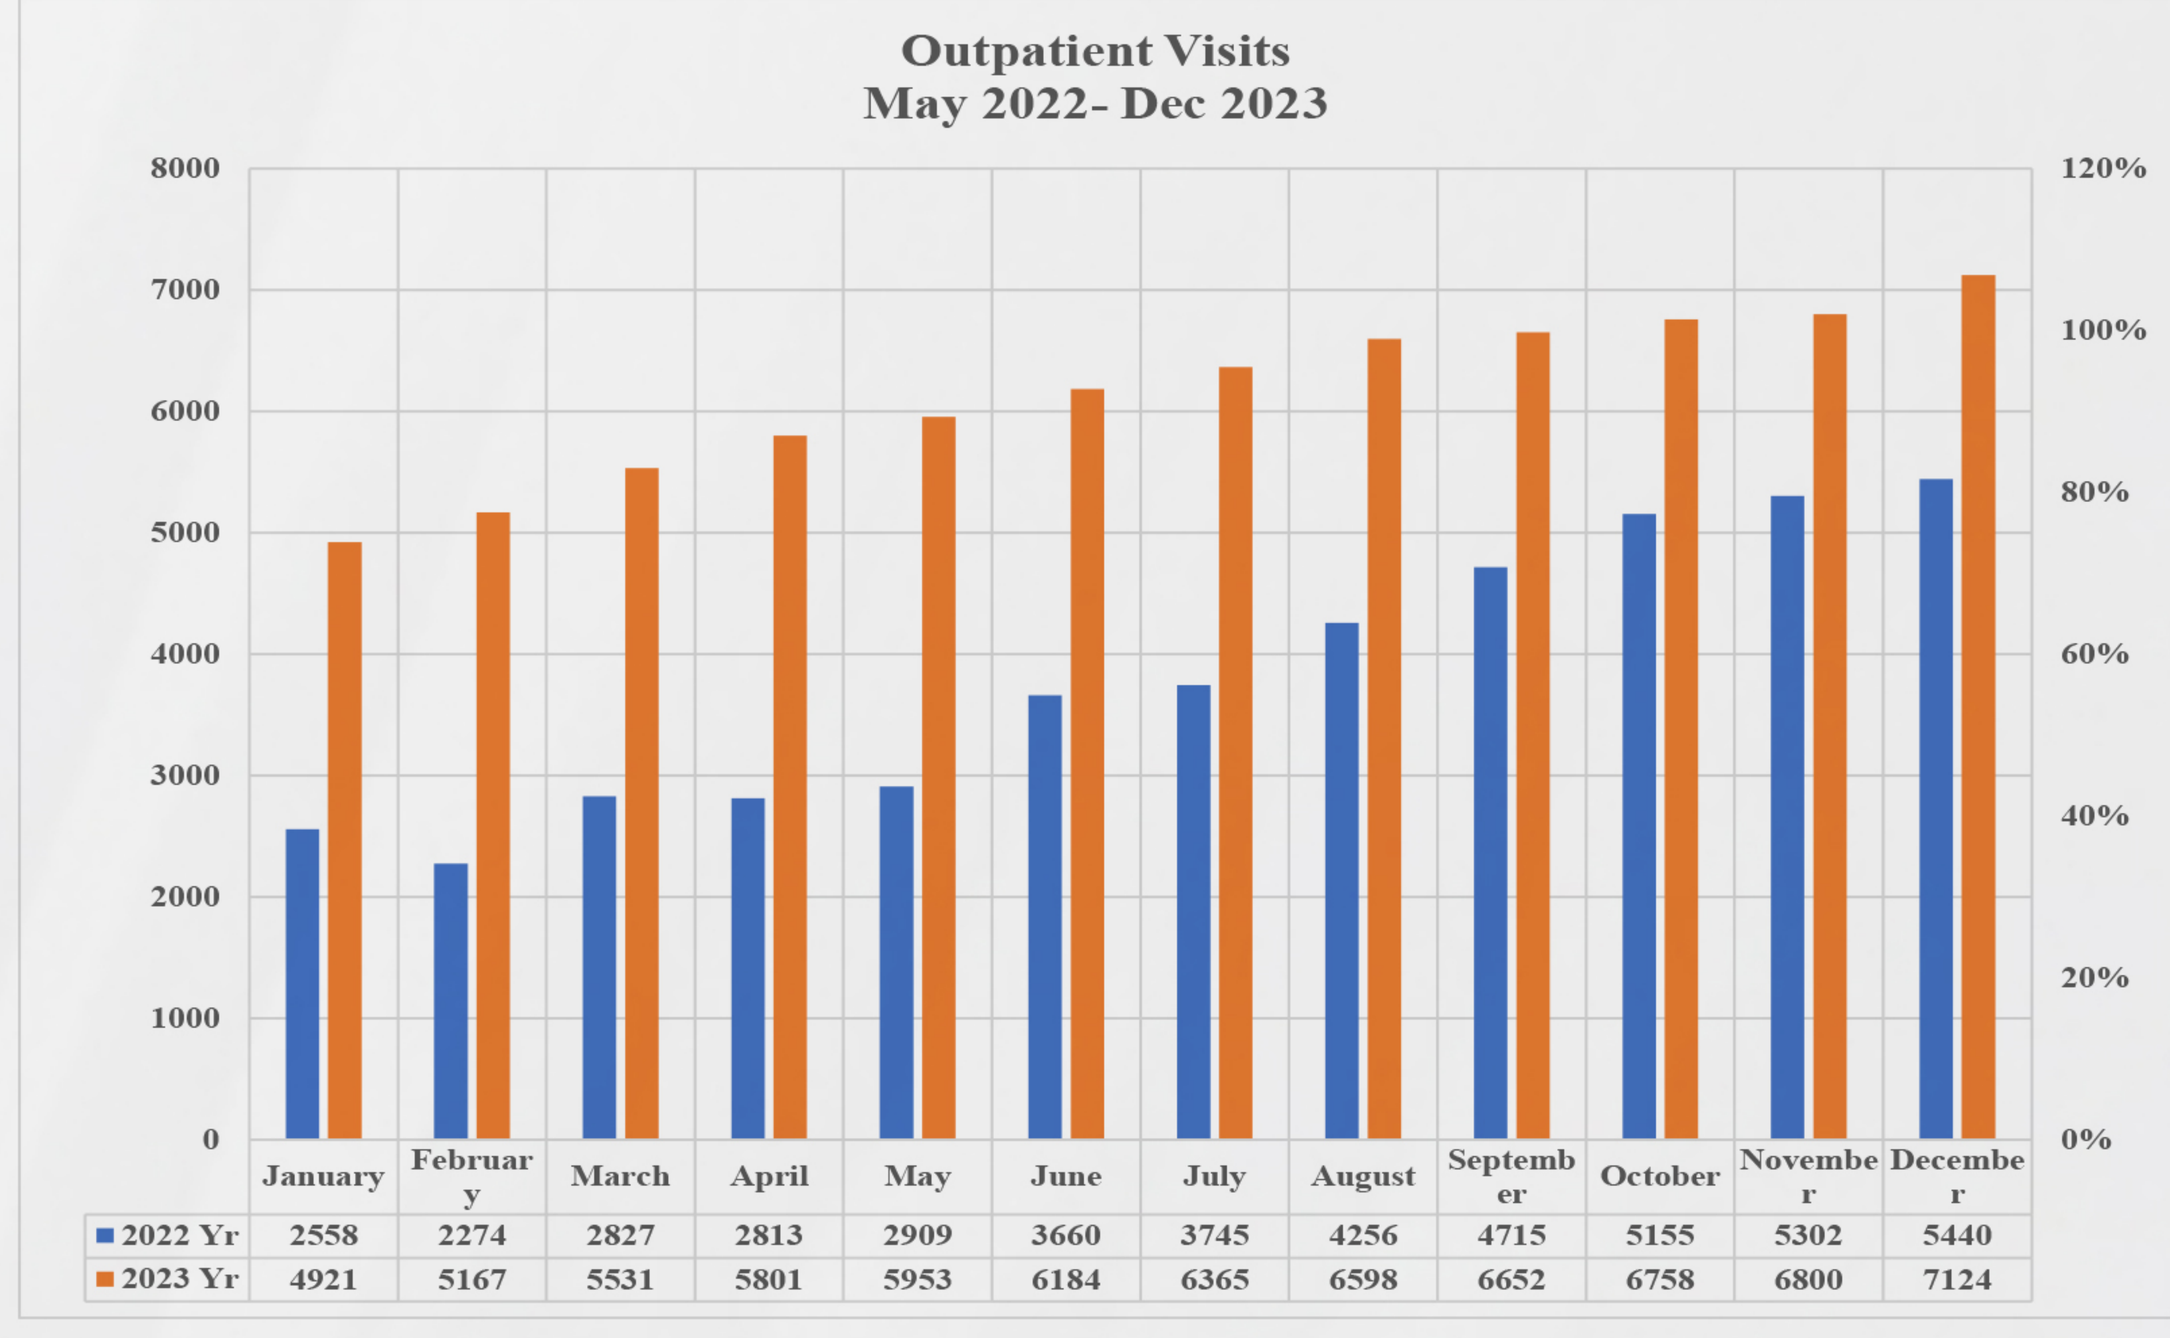
Outpatient Visits
May 2022- Dec 2023

	January	February	March	April	May	June	July	August	September	October	November	December
2022 Yr	2558	2274	2827	2813	2909	3660	3745	4256	4715	5155	5302	5440
2023 Yr	4921	5167	5531	5801	5953	6184	6365	6598	6652	6758	6800	7124

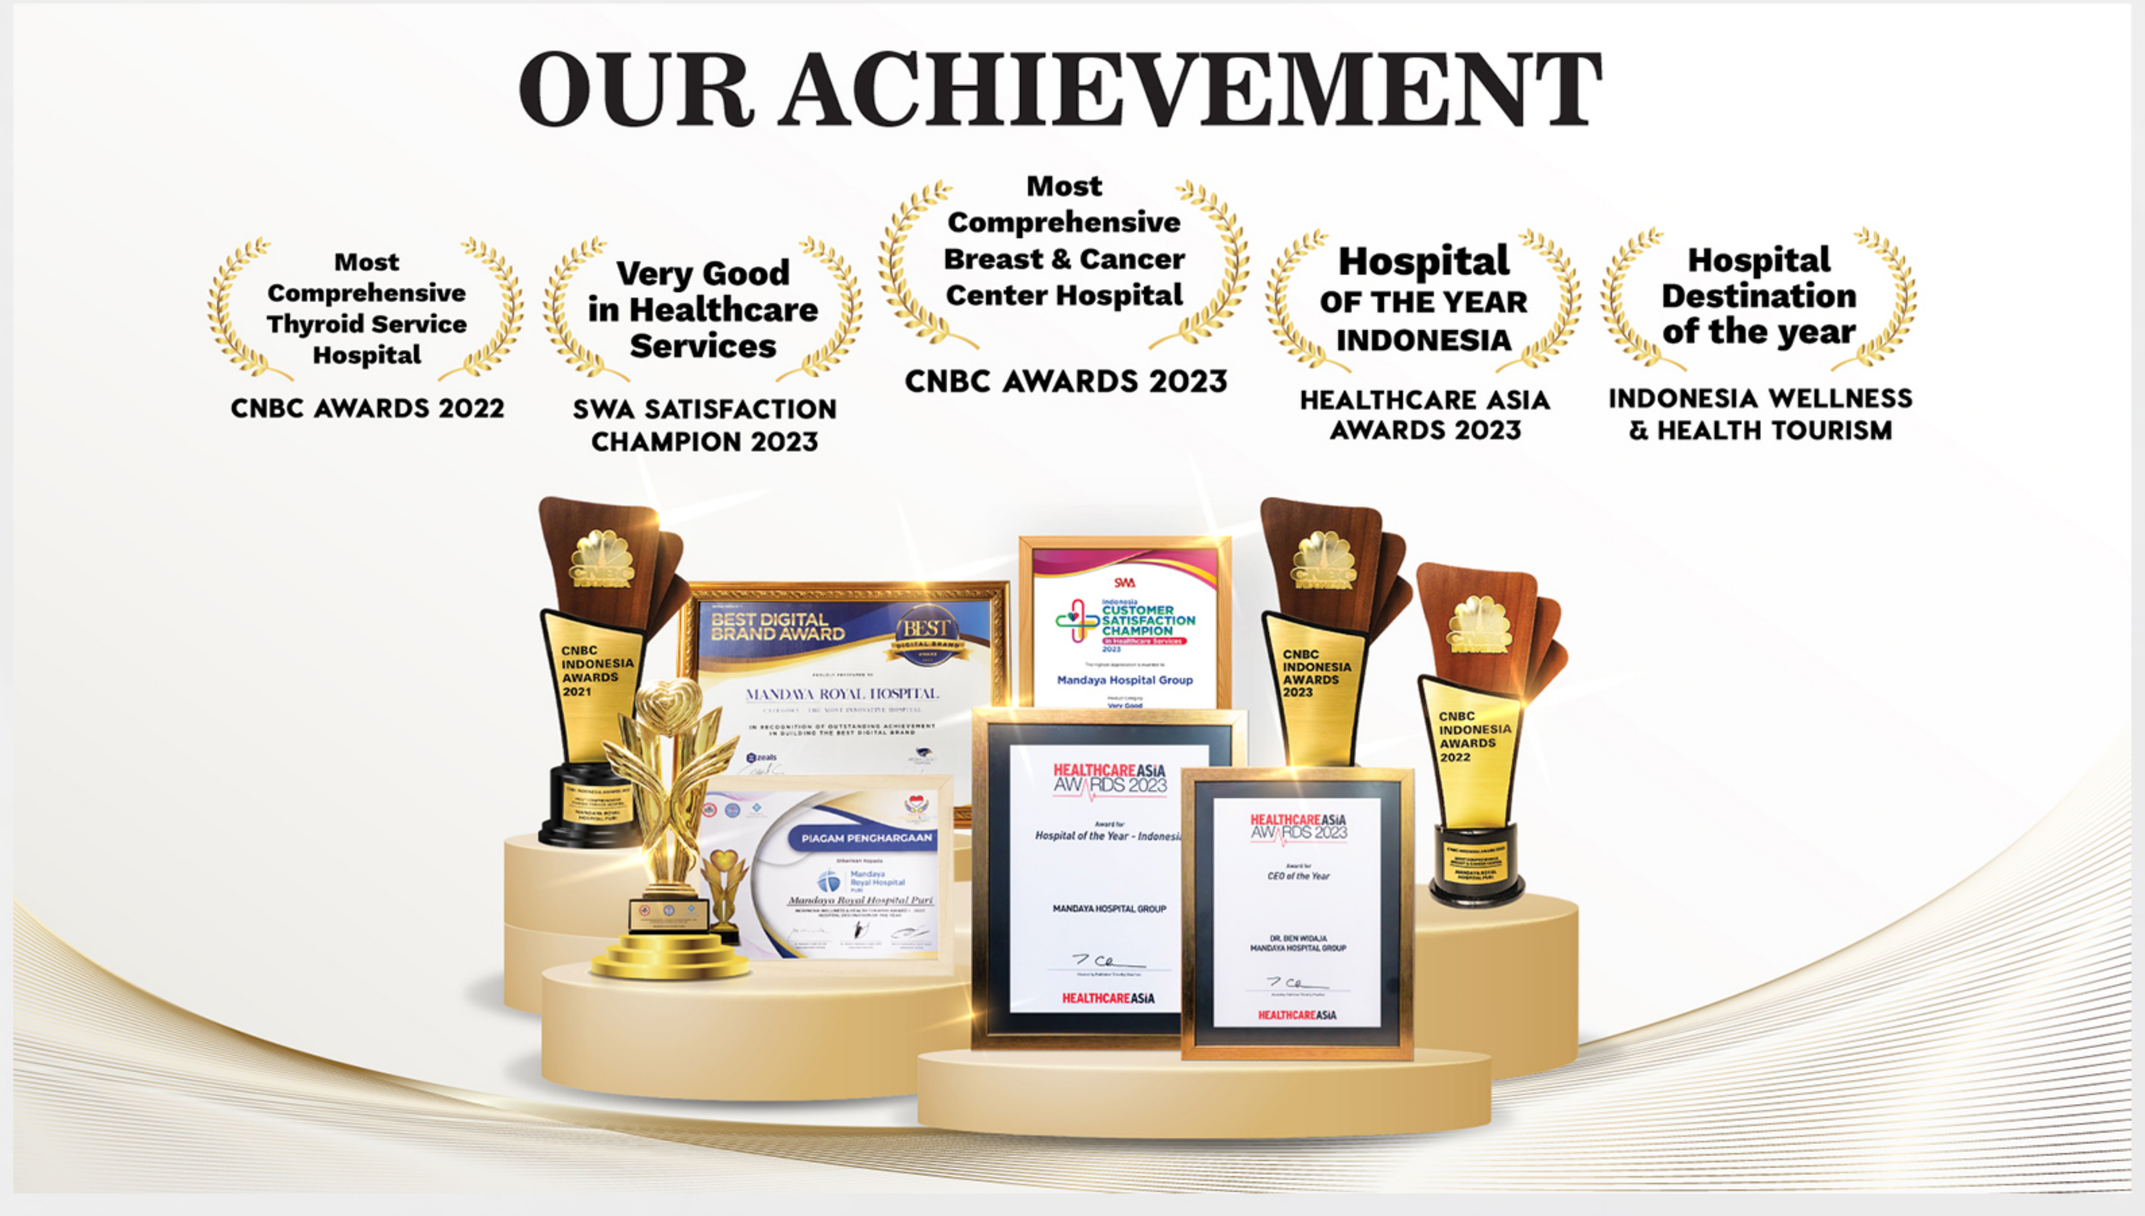
OUR ACHIEVEMENT
Most Comprehensive Thyroid Service Hospital
CNBC AWARDS 2022
Very Good in Healthcare Services
SWA SATISFACTION CHAMPION 2023
Most Comprehensive Breast & Cancer Center Hospital
CNBC AWARDS 2023
Hospital OF THE YEAR INDONESIA
HEALTHCARE ASIA AWARDS 2023
Hospital Destination of the year
INDONESIA WELLNESS & HEALTH TOURISM

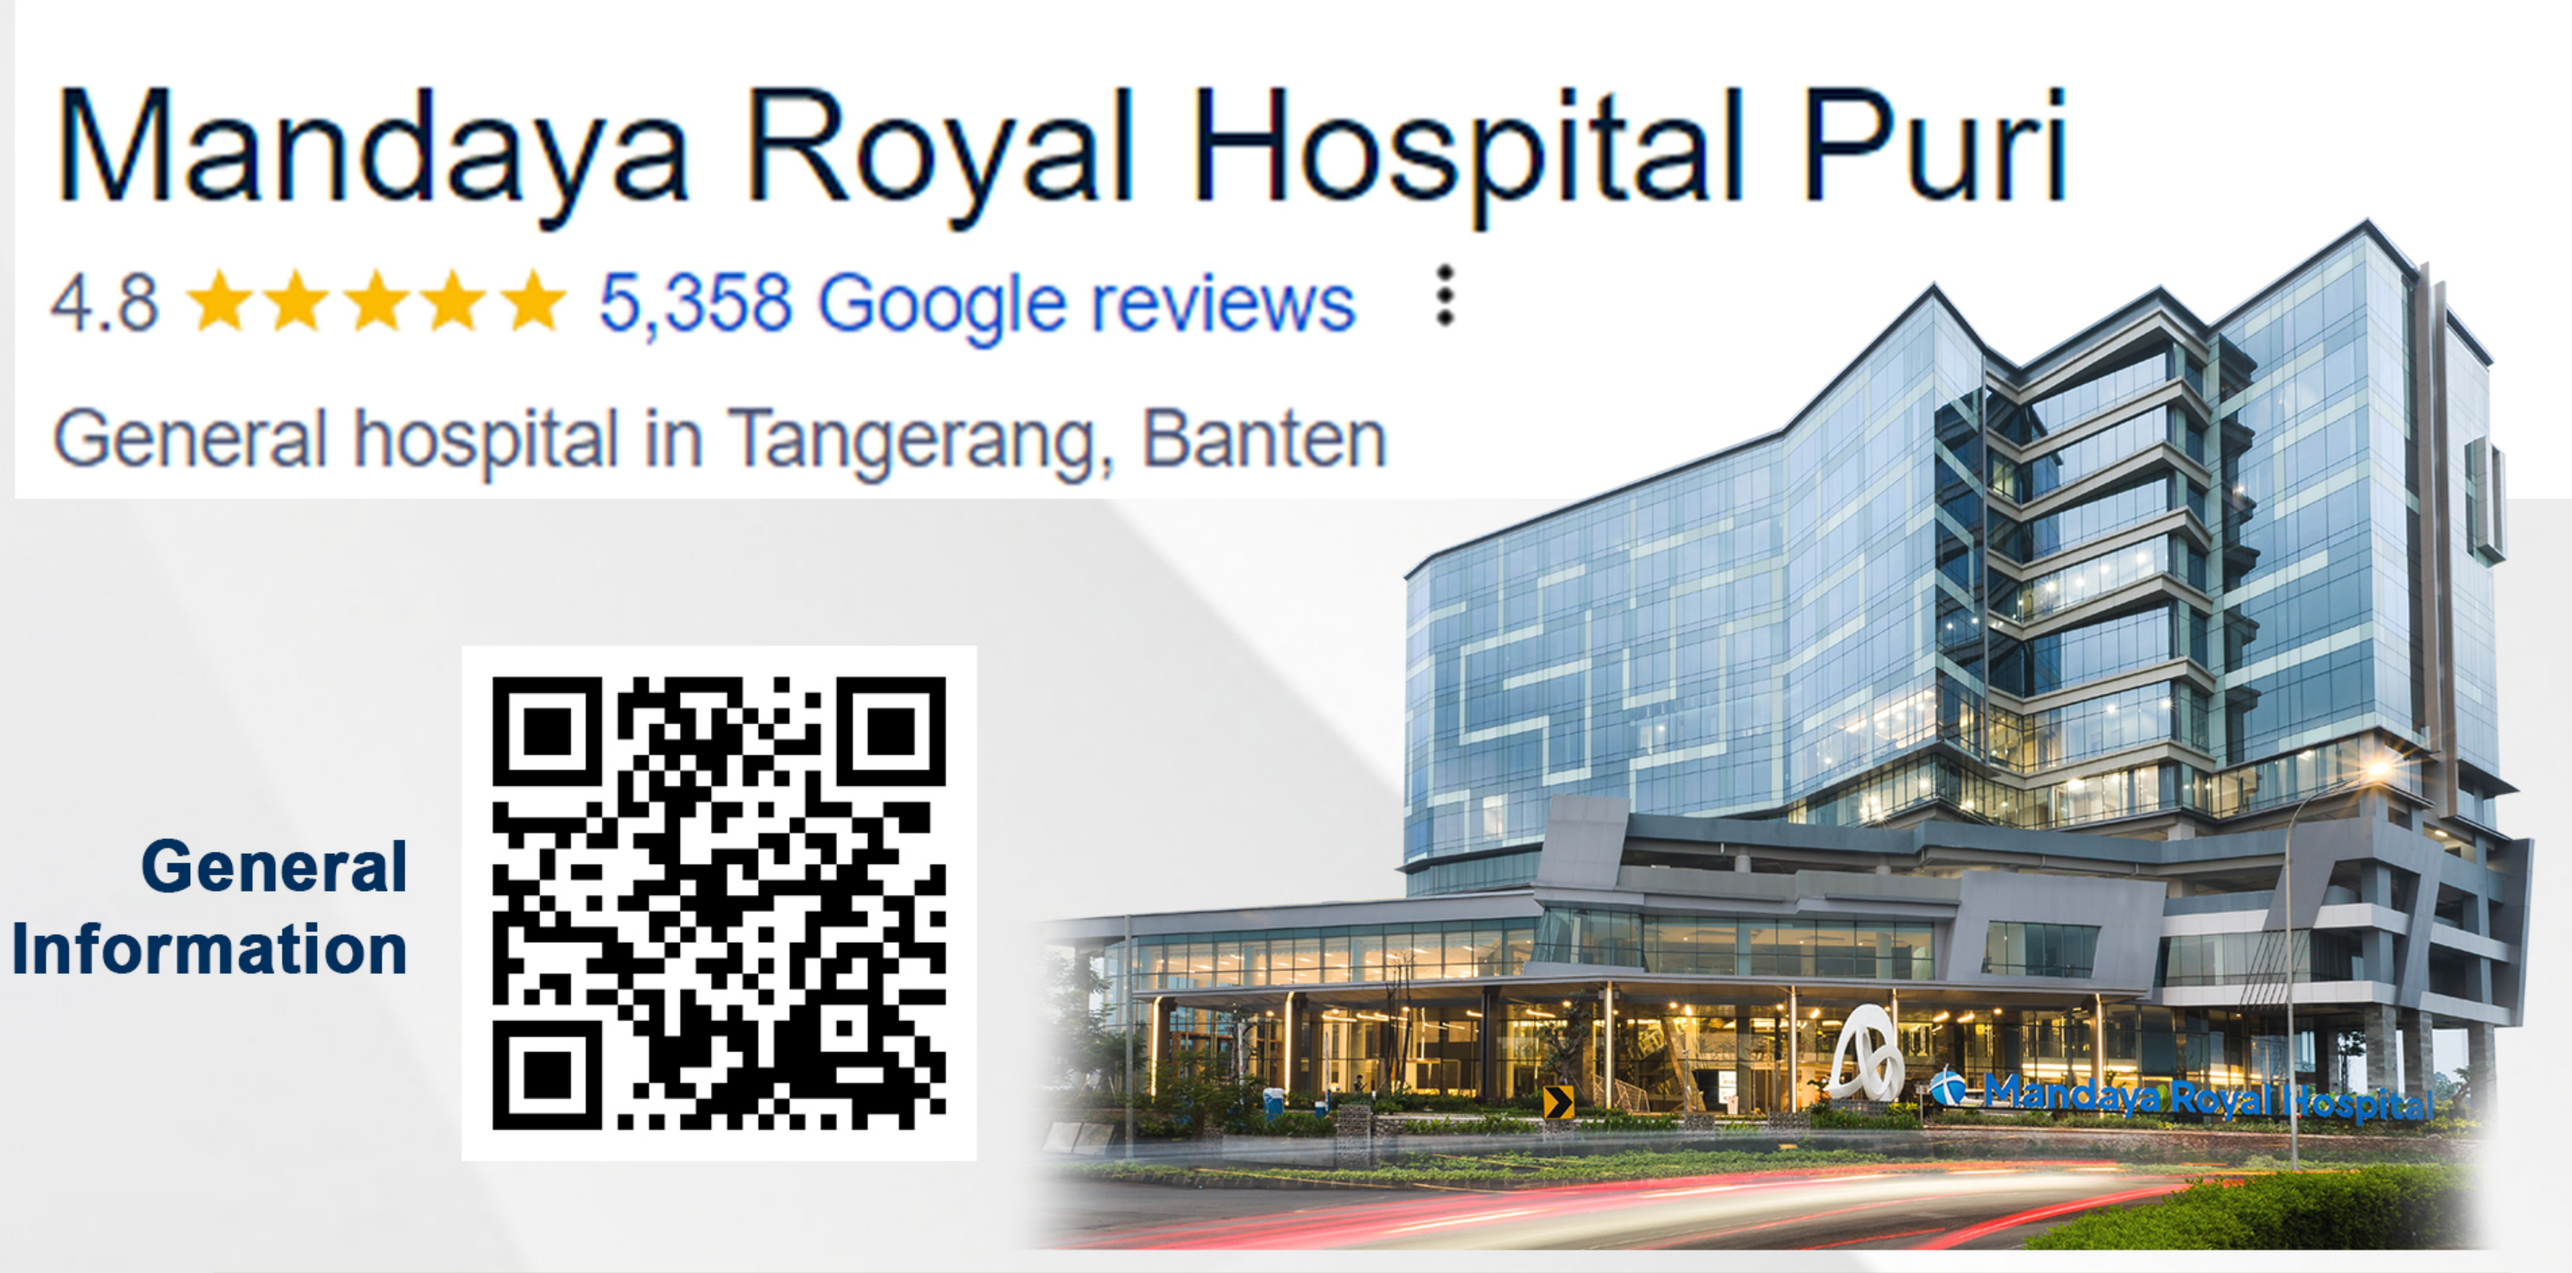
Mandaya Royal Hospital Puri
4.8 ★★★★★ 5,358 Google reviews
General hospital in Tangerang, Banten
General Information

CONCLUSIONS

1) The patient's experience of the health care system and caring while being treated at the Mandaya Royal Hospital is generally positive.
2) Patient-family and community-centered care models is an important step that causes healthcare workers to look beyond the physical condition of the person being treated; allows the patient as a person to be brought to the forefront and allows for their individual needs to be met and for care to be enhanced.
3) The Planetree patient-centered model of care positively affected patient satisfaction, length of stay, and cost per case.

REFERENCES

1) Planetree International. Person-Centered care certification program manual. Planetree International, 2017
2) Doyle C, Lennox L, Bell D. A systematic review of evidence on the links between patient experience and clinical safety and effectiveness. BMJ Open 2013;3:e001570.
3) World Health Organization. People-centred health care: a policy framework. World Health Organization, 2007.

ACKNOWLEDGE

Praise and gratitude we pray to the presence of God Almighty. We express our deepest appreciation to all parties in Mandaya Royal Hospital Puri which provides the opportunity to conduct the project in this hospital. Indonesian Commission on Accreditation of Hospitals, and our beloved family who always support us to take an active role in ISQua events. This project is still far away from perfect, but it is hoped that it will be useful not only for hospital, but also for readers.

CONTACT INFORMATION

HEART & VASCULAR CENTER · Brave Breast Advanced Cancer Center · ADVANCED CANCER CENTER · Orthopedic & Robotic Center · Mandaya Neuromuscular Center · Cognitive & Sleep Clinic · BRAIN-SPINE-PAIN CENTER · ADVANCED CANCER & RADIOTHERAPY CENTER · Pusat Tiroid Indonesia by Mandaya Royal Hospital

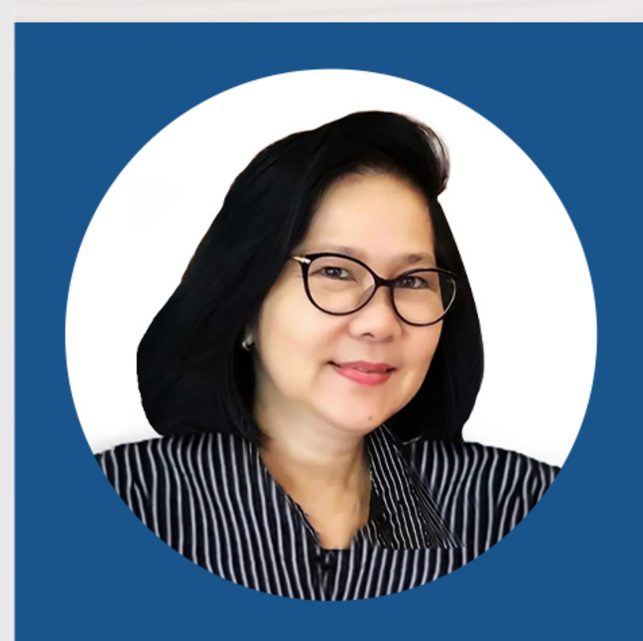

: +62 816 1982 736
: CEO Mandaya Hospital Group
: annatahjoo@gmail.com