مستشفى الولادة والأطفال بالدمام
Maternity And Children Hospital - Dammam
Eastern Health Cluster تجمع الشرقية الصحي

ISQUA2024-ABS-2753

Integrating Cervical Cancer Screening Program as Modle of Care in Eastern Health Cluster

Zahra A. Al-Turaifi[1], Muna Alshehabi[2], Dalia Al-OUF[1]

1* Maternity and children hospital , Dammam , EHC 2* Modle of care, Dammmam, EHC

introduction

Cervical cancer is a major global health burden, especially for women who face significant challenges due to limited resources and limited access to healthcare services. The Pap smear and cervical cancer screening programs are essential for the early detection and prevention of the disease. This poster discusses the implementation and effectiveness of Pap smear-based cervical cancer screening programs in Saudi Arabia's infrastructure screening program, as well as the associated barriers and potential solutions.

Methods

The development of an effective cervical cancer screening program involves various key analysis and engagement activities within EHC.

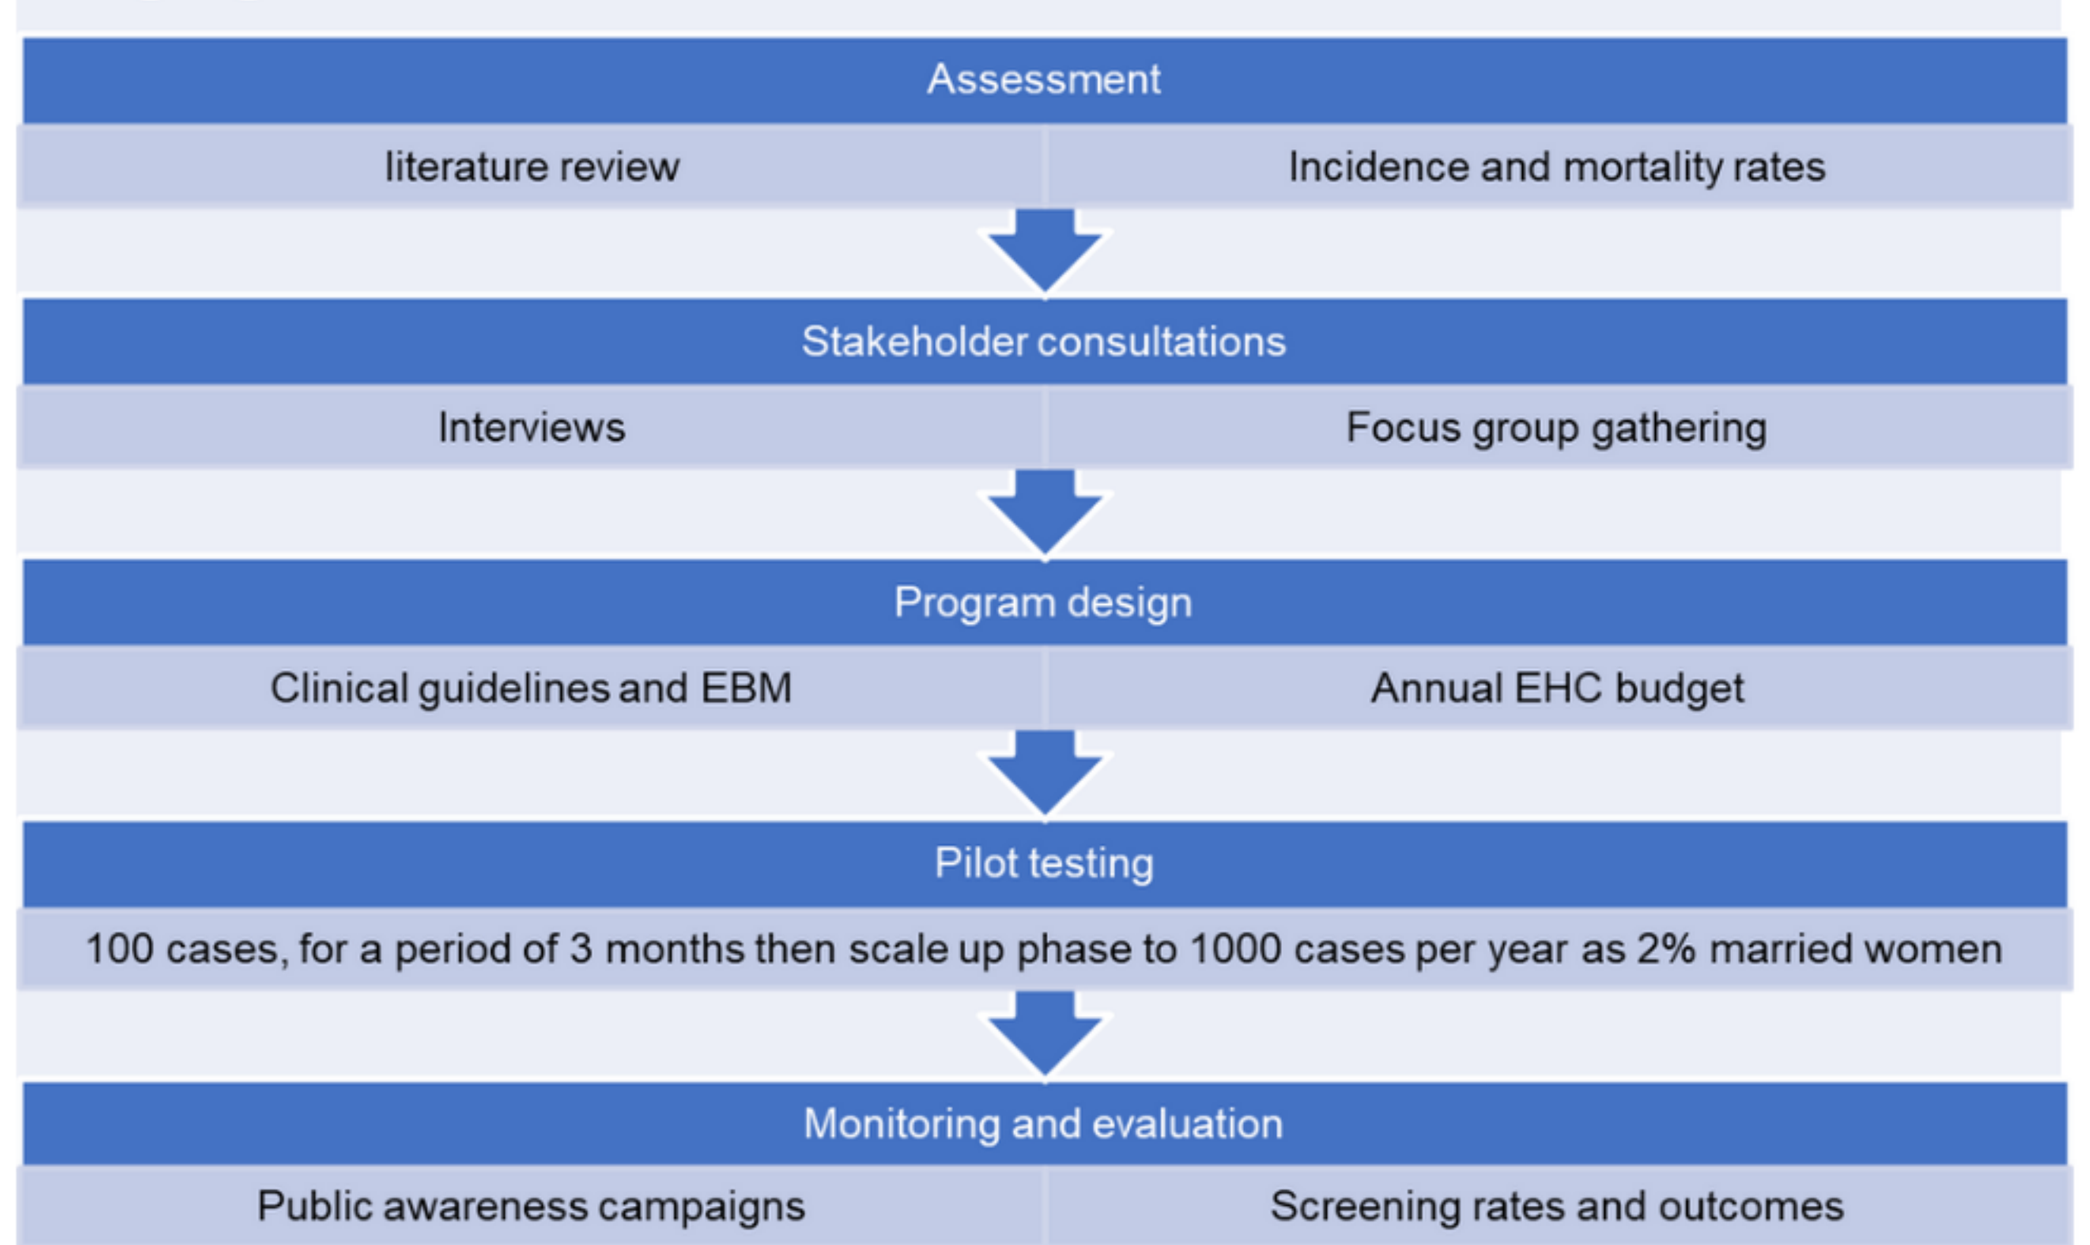

Assessment
literature review
Incidence and mortality rates
Stakeholder consultations
Interviews
Focus group gathering
Program design
Clinical guidelines and EBM
Annual EHC budget
Pilot testing
100 cases, for a period of 3 months then scale up phase to 1000 cases per year as 2% married women
Monitoring and evaluation
Public awareness campaigns
Screening rates and outcomes

By following these steps, a comprehensive and effective cervical cancer screening program was developed and established .

Results

The performance and changes in this screening program, according to that several approaches implemented. Continuous Quality Improvement (CQI): data analysis, and feedback mechanisms in identify areas for improvement. (KPIs) (fig.1) These can be demonstrated through improved screening rates and increased adherence to recommended follow-up procedures.

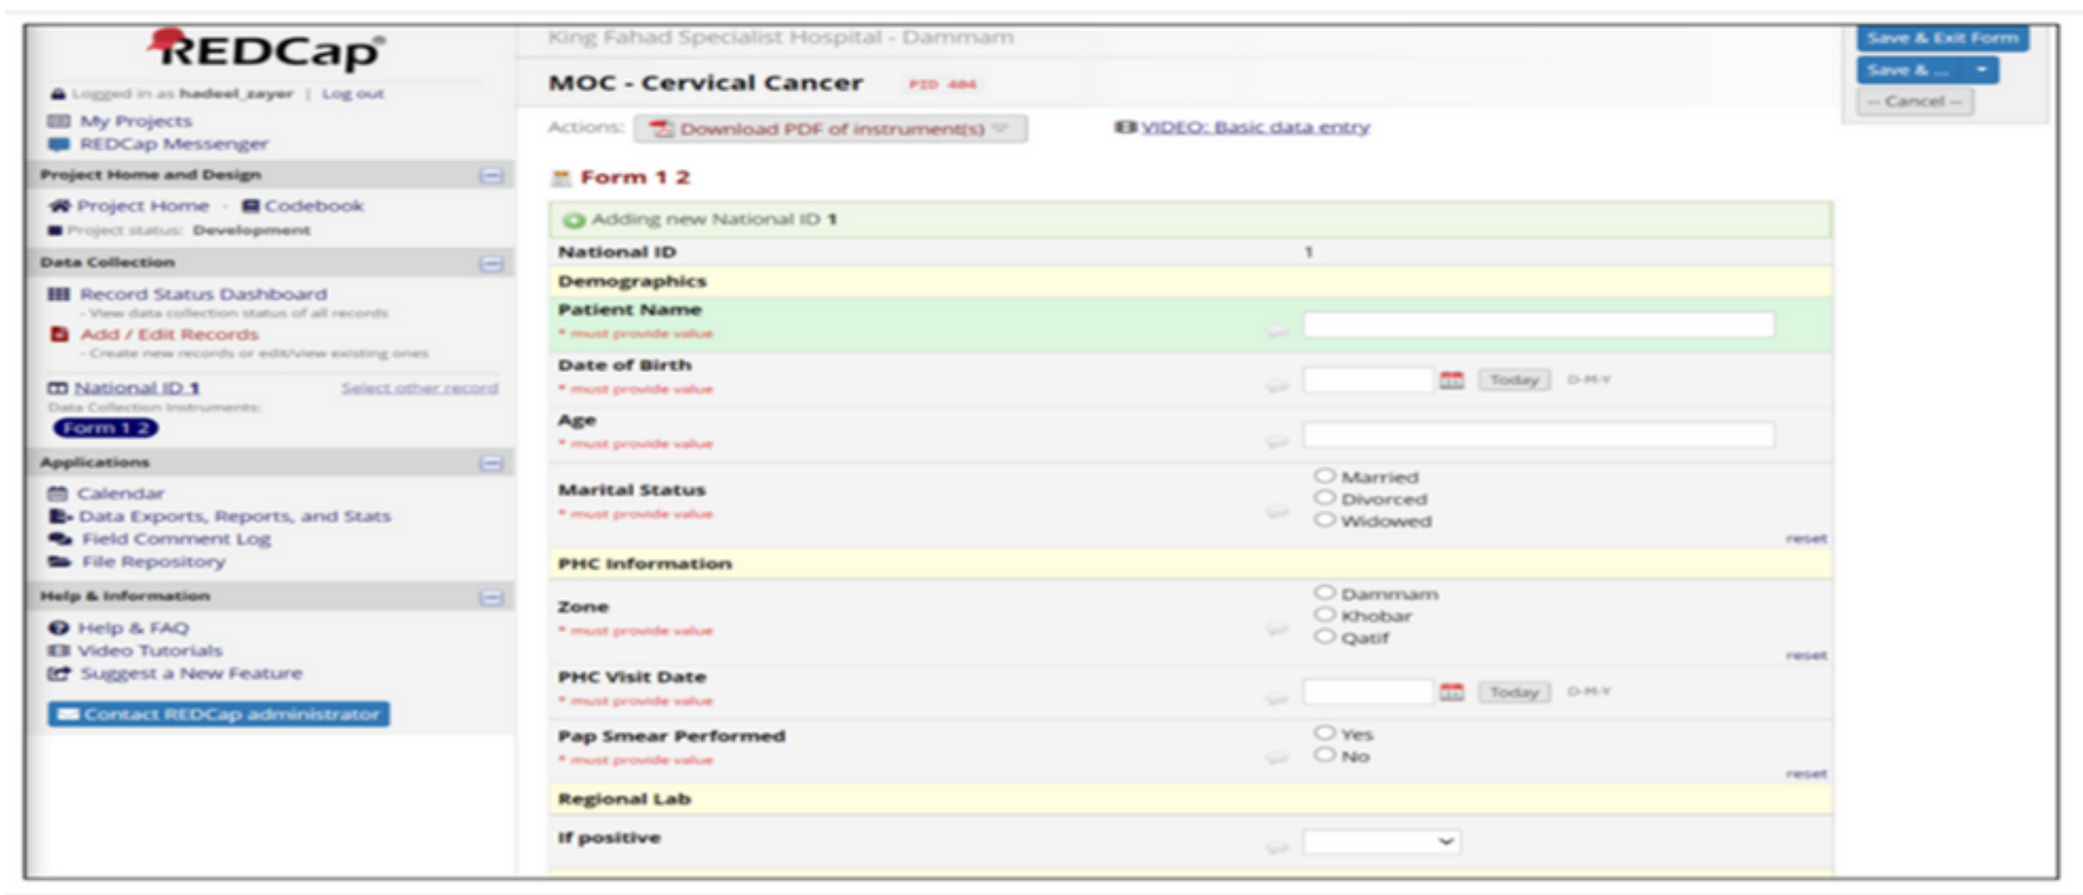

REDCap
MOC - Cervical Cancer

(fig.1: Redcap software-entering screen)

Implementing systematic recall and reminder systems was effective strategy to encourage timely and regular screening facilitated by care coordinators. These KPIs was calculated through RedCap which used in evaluating this program include the percentage of pap smears (fig.2), average wait time from pap smear to lab result, percentage of pap smear rejections, average wait time from lab result to primary healthcare receipt of results, (fig.3)

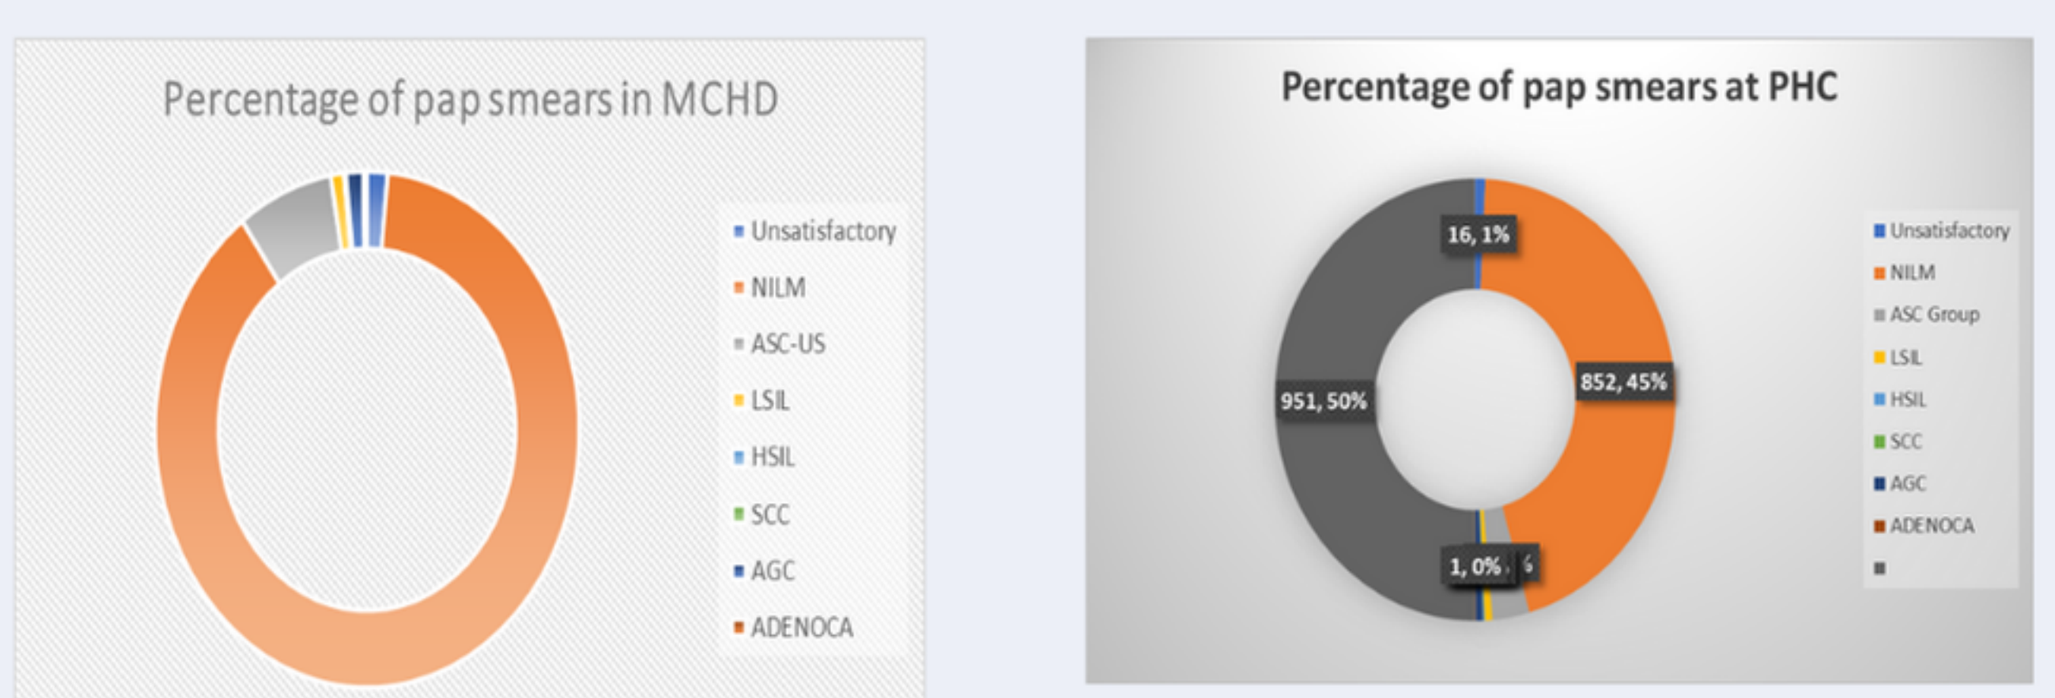

Percentage of pap smears in MCHD
Unsatisfactory
NILM
ASC-US
LSIL
HSIL
SCC
AGC
ADENOCA
Percentage of pap smears at PHC
16, 1%
951, 50%
852, 45%
1, 0%
Unsatisfactory
NILM
ASC Group
LSIL
HSIL
SCC
AGC
ADENOCA

Fig2: Percentage of pap smears MCHD vs PHC

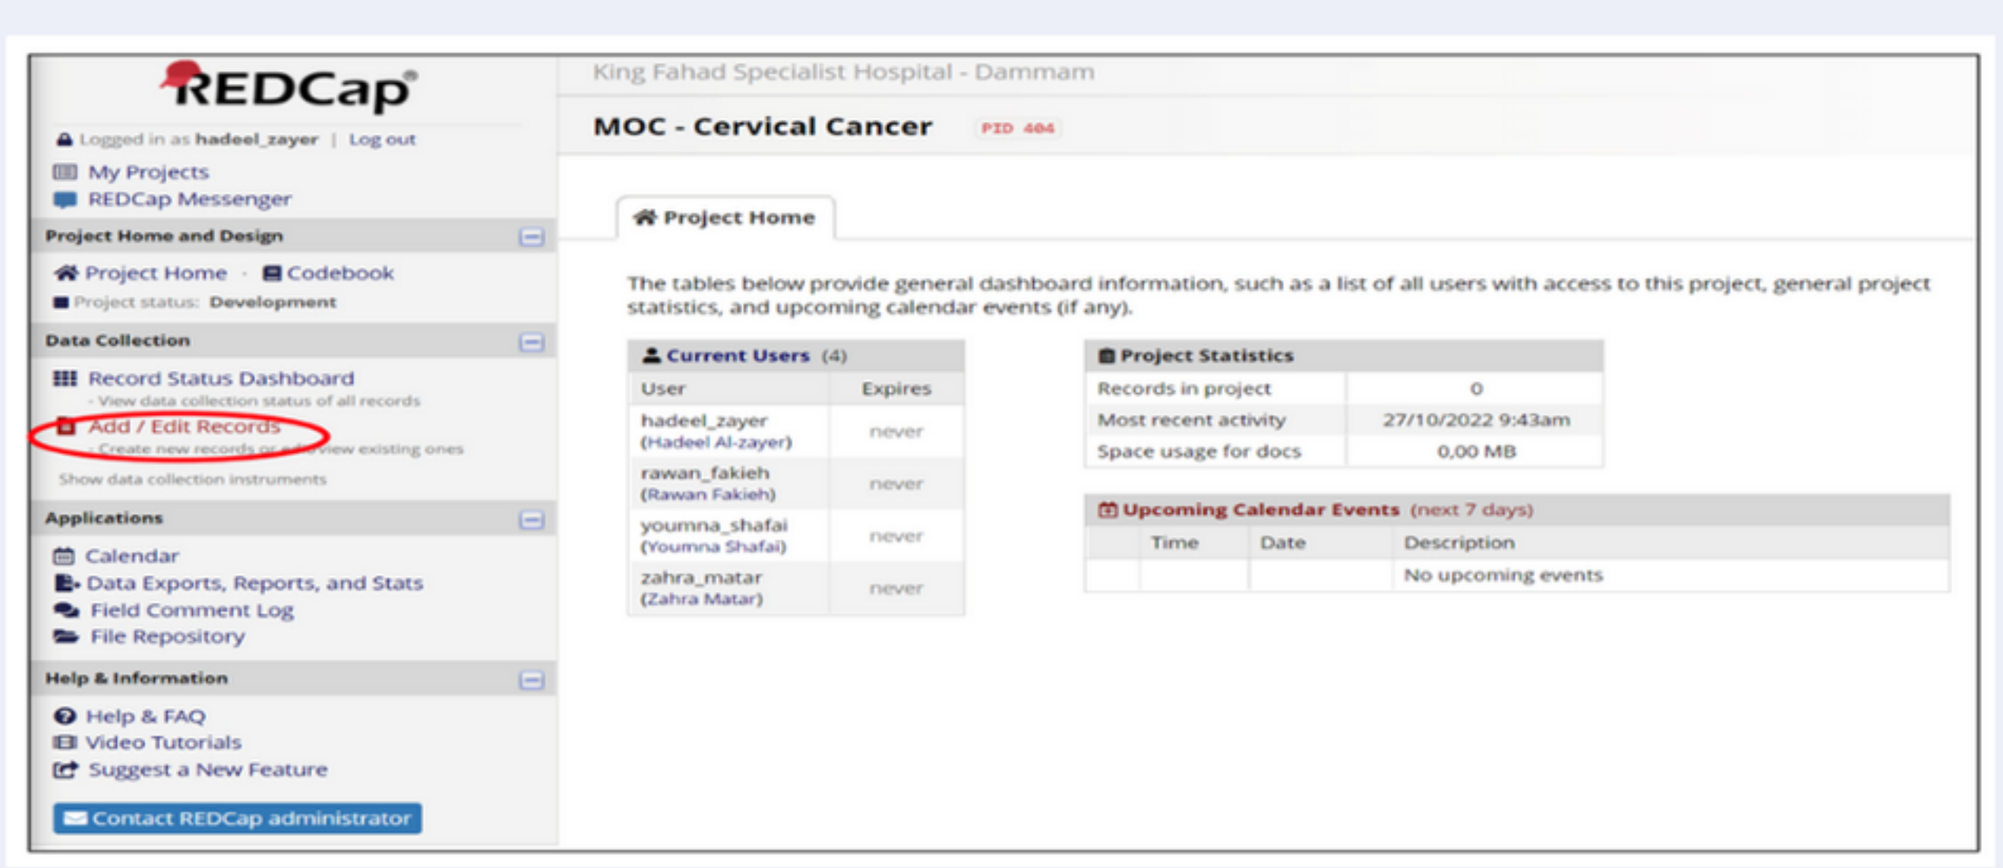

REDCap
MOC - Cervical Cancer
Project Home

fig.3-1: Redcap software (Extraction data screen)

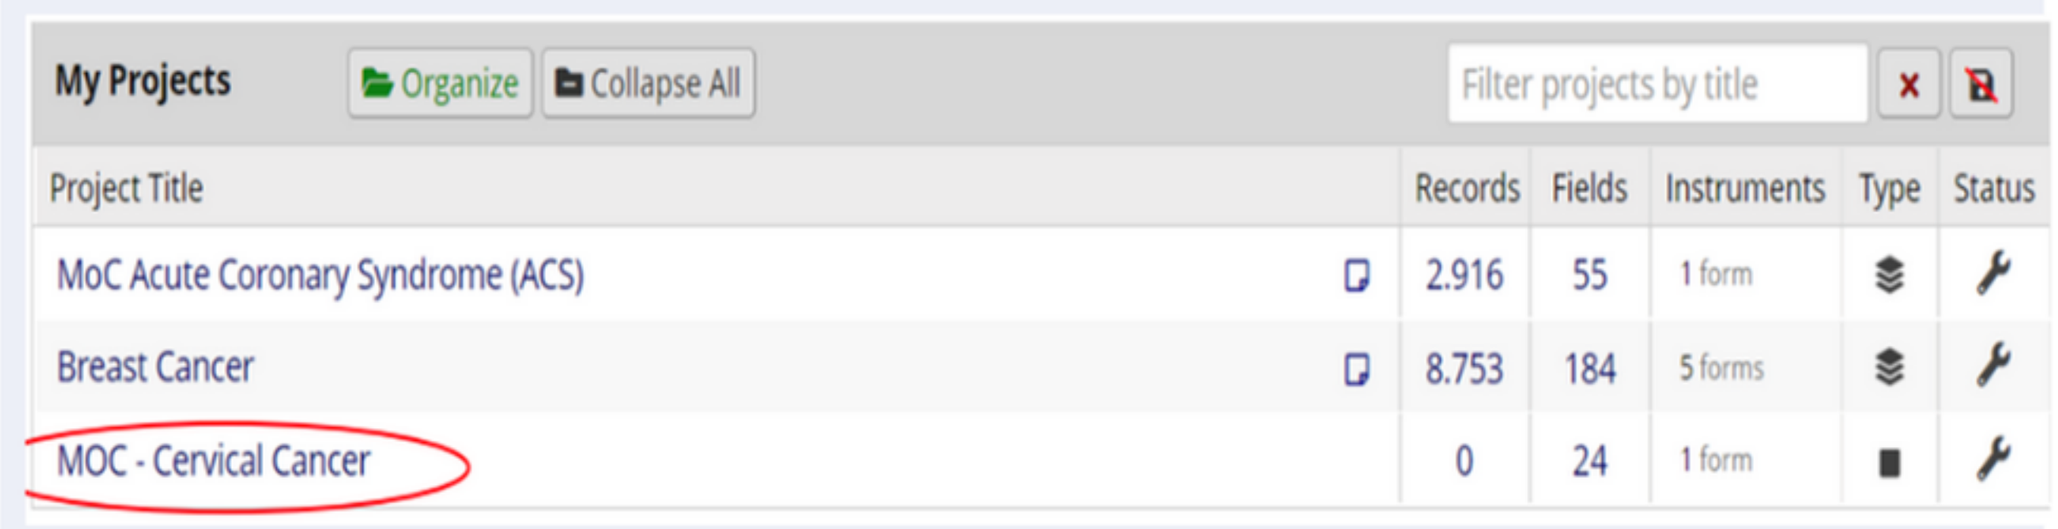

My Projects
Project Title
Records
Fields
Instruments
MoC Acute Coronary Syndrome (ACS)
Breast Cancer
MOC - Cervical Cancer

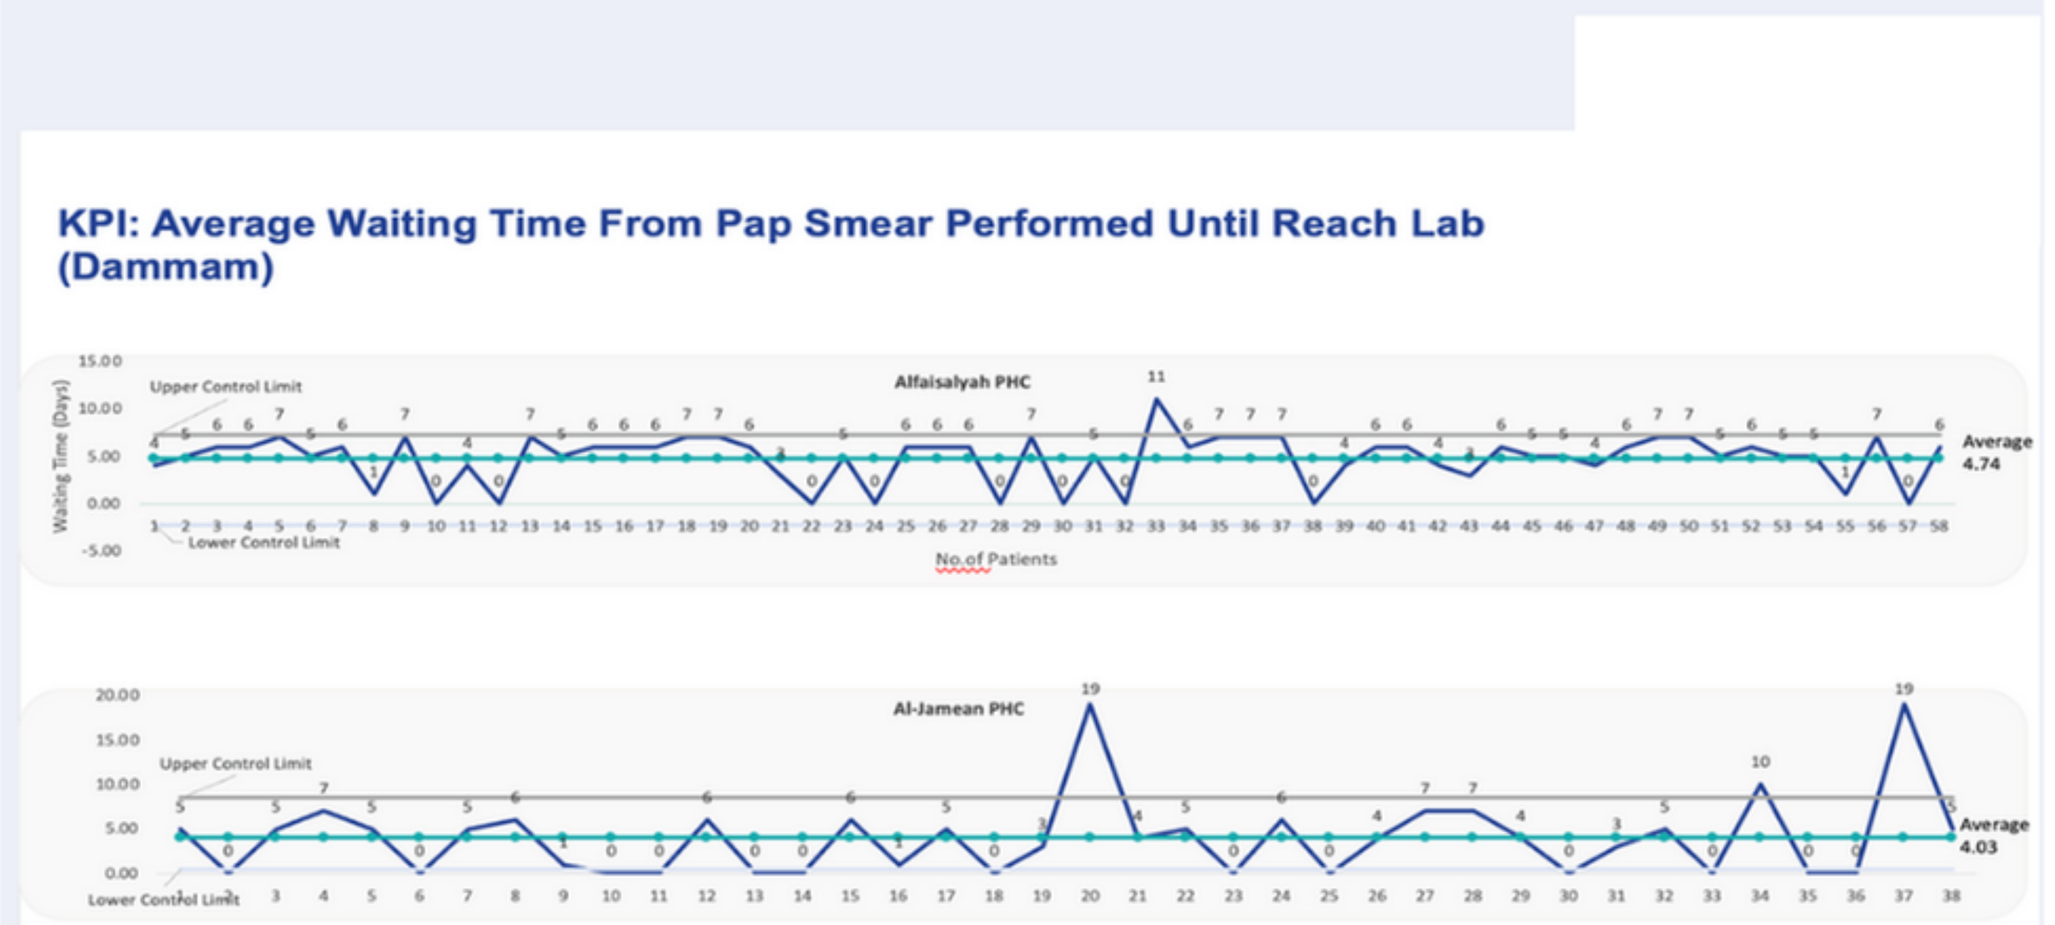

KPI: Average Waiting Time From Pap Smear Performed Until Reach Lab (Dammam)
Alfaisalyah PHC
Al-Jamean PHC
Upper Control Limit
Lower Control Limit
Average 4.74
Average 4.03

Fig.3-2: Average wait time from lab result to primary healthcare receipt of results

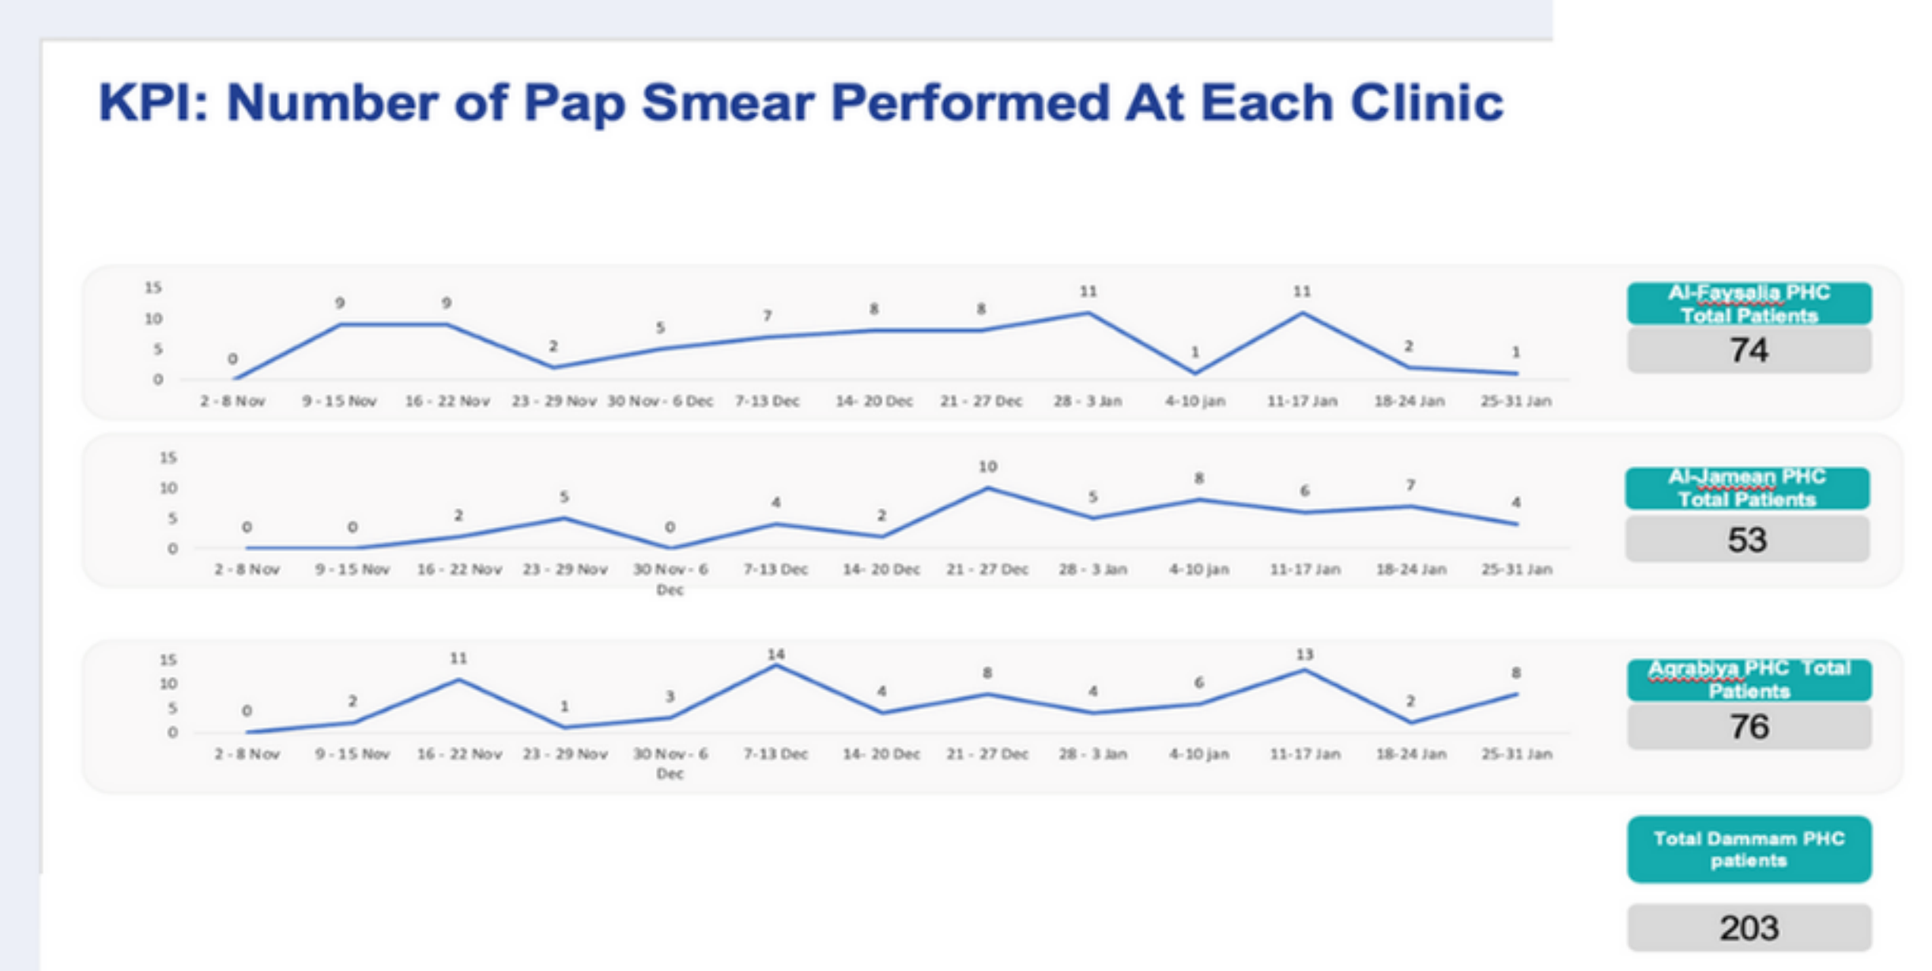

KPI: Number of Pap Smear Performed At Each Clinic
74
53
76
Total Dammam PHC patients
203

Fig.3-3: Number of PAP smear performed at each clinic

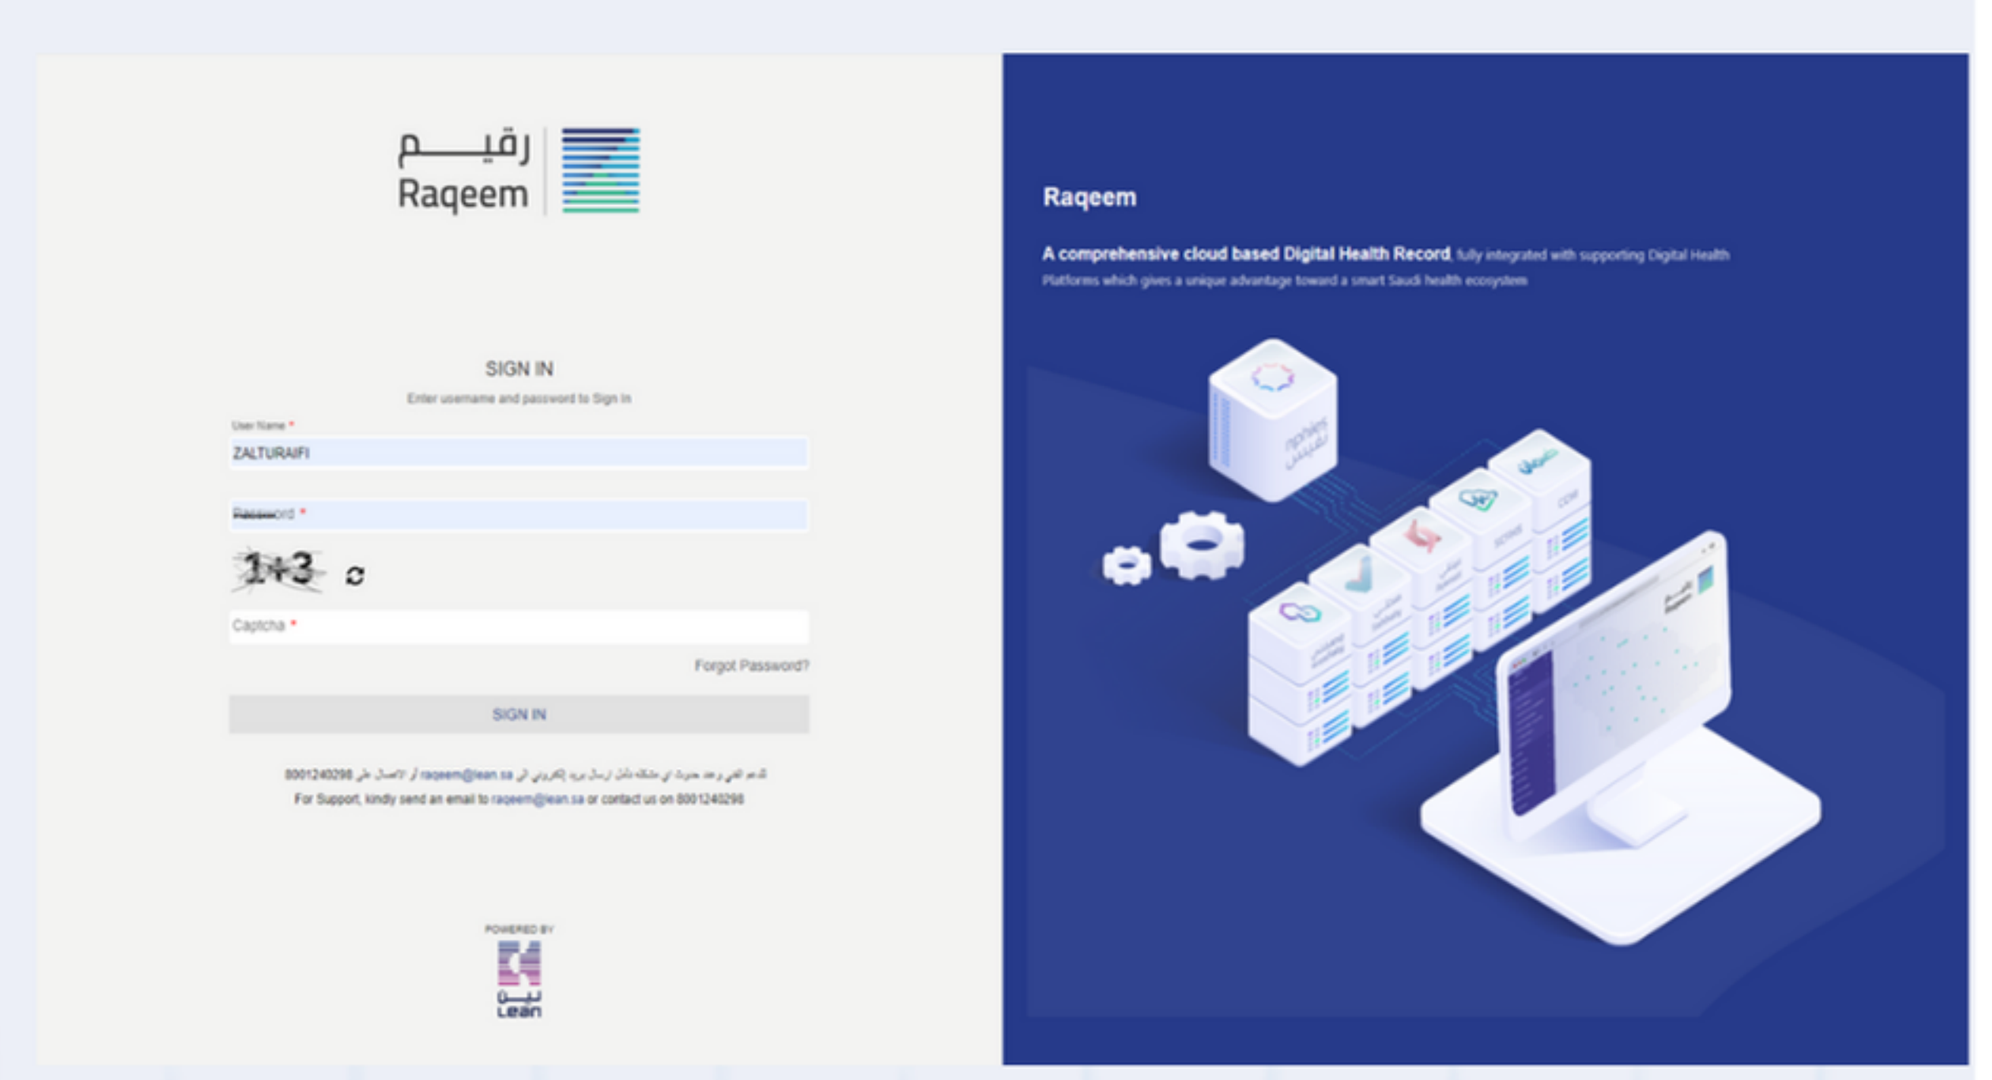

رقيم
Raqeem
SIGN IN
Raqeem

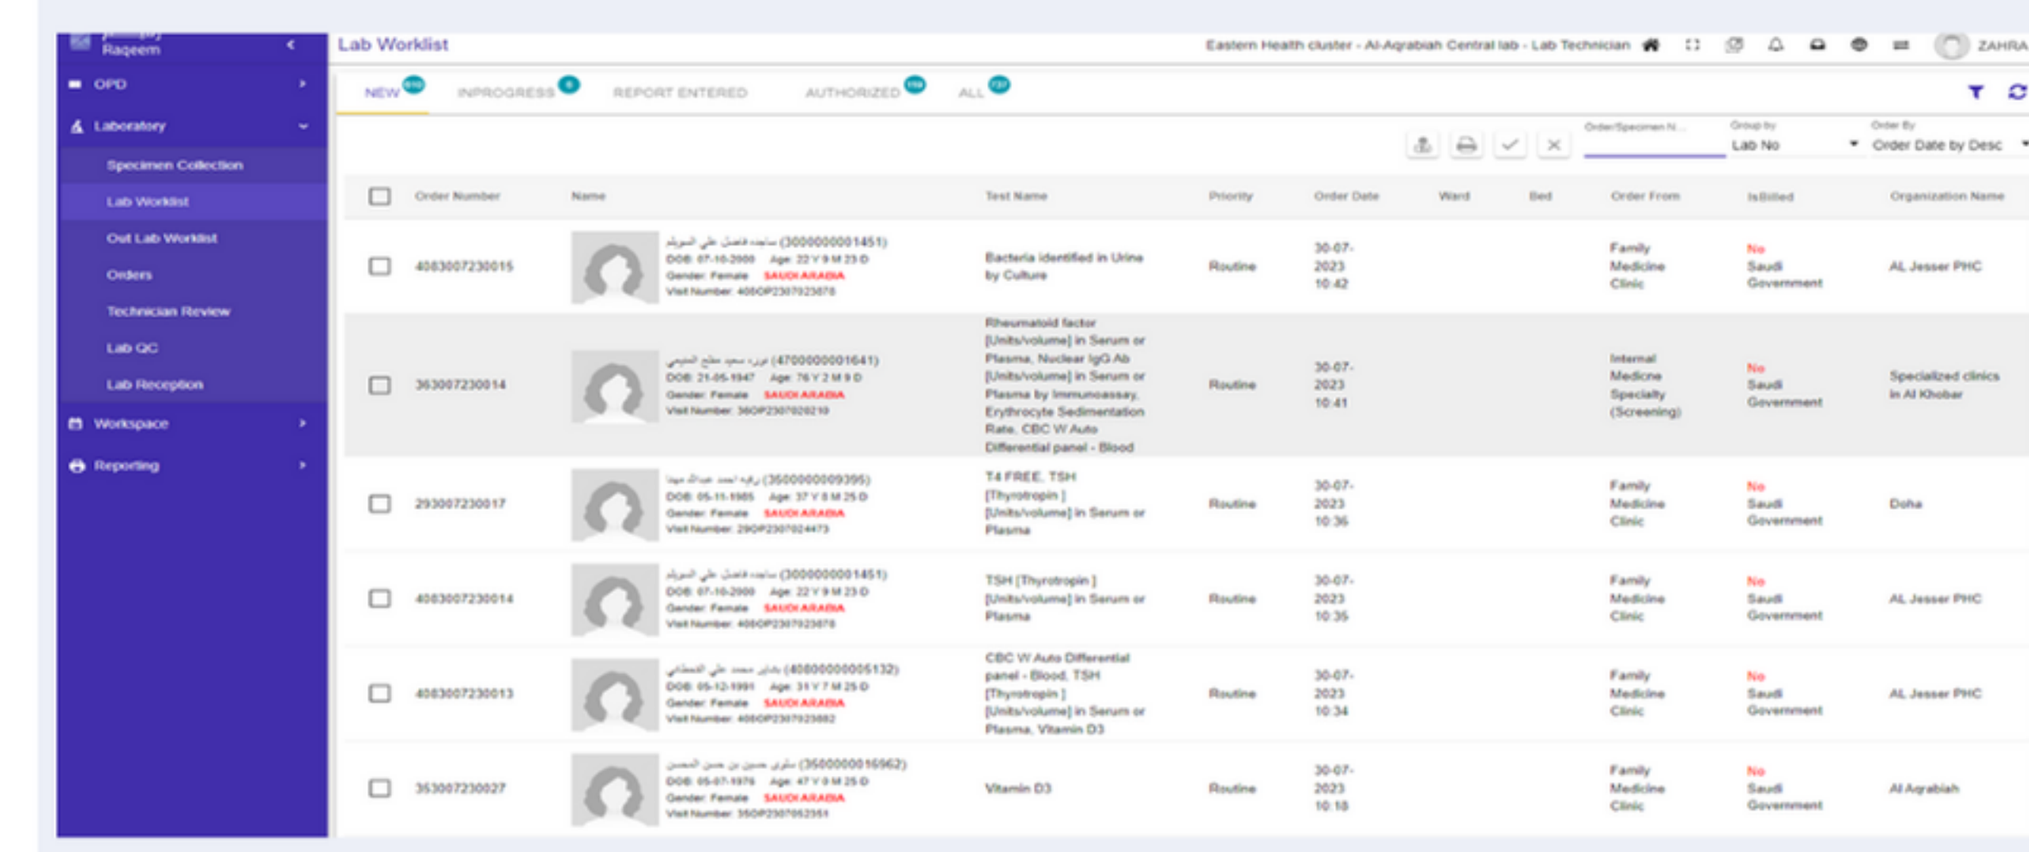

Lab Worklist

Fig.3-4: Raqeem software (Connect system between laboratory hospital and PHCs doctors and lab to dispatch the final results)

average wait time from a positive result to referral to a tertiary hospital, percentage of time between secondary and tertiary hospital referrals, percentage of patients with positive cervical cancer results, and cervical cytology classification rates based on CAP classification rate percentile as a benchmark (fig.4).

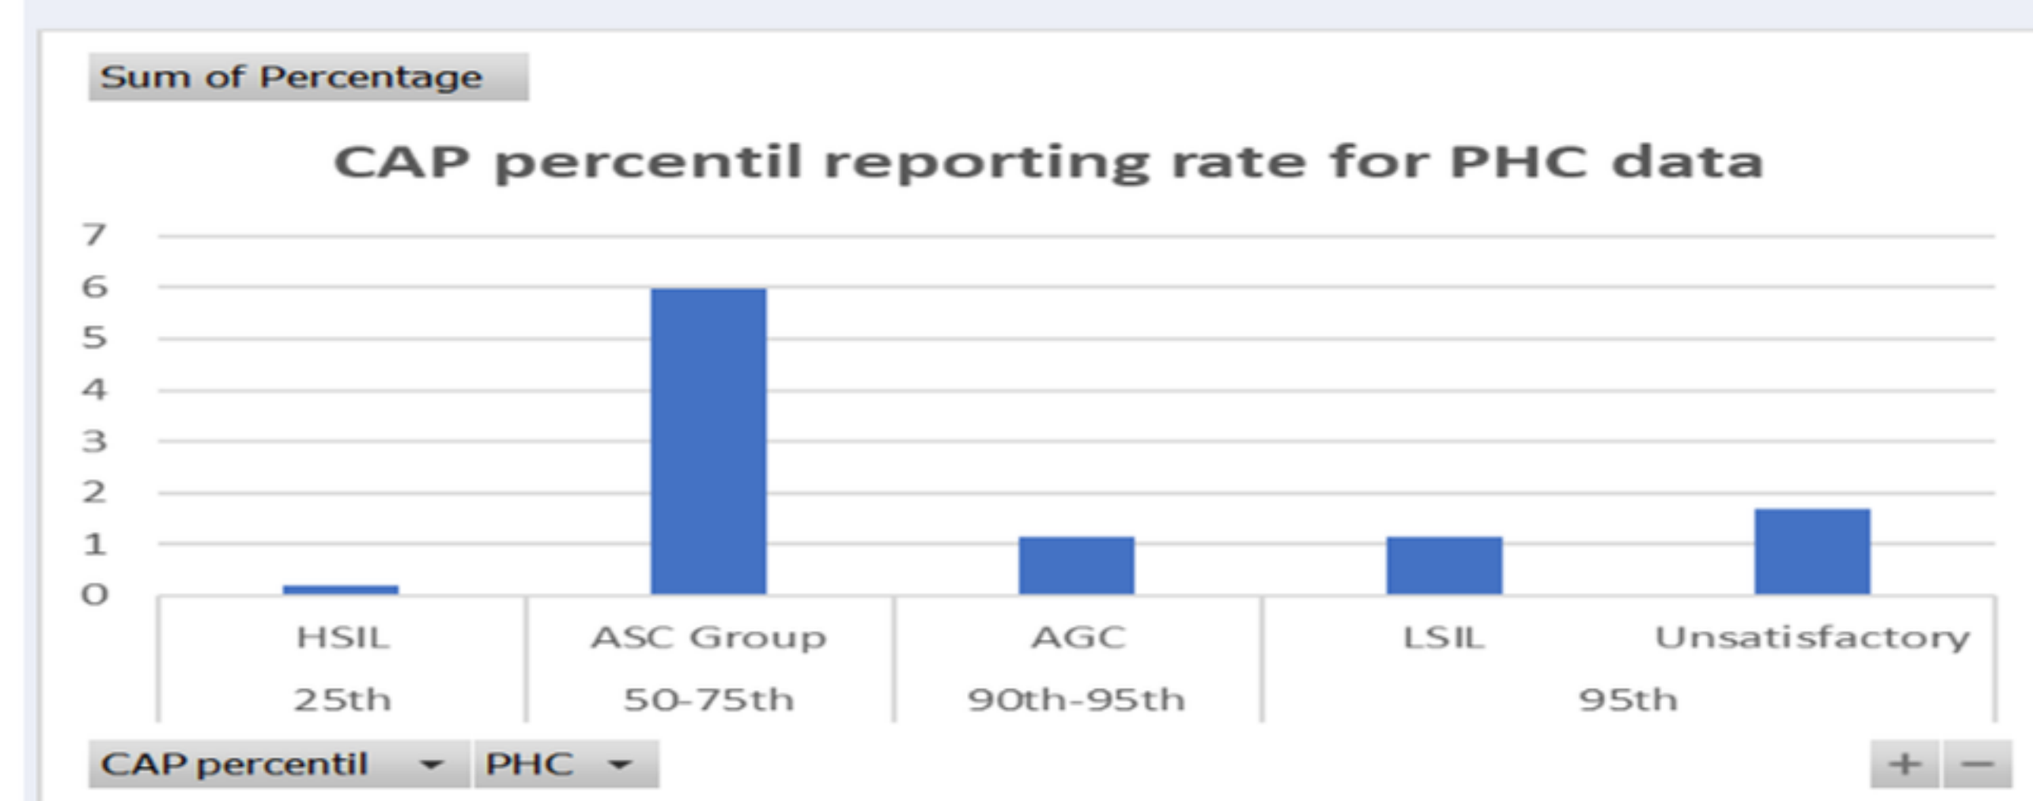

Sum of Percentage
CAP percentil reporting rate for PHC data
HSIL
25th
ASC Group
50-75th
AGC
90th-95th
LSIL
Unsatisfactory
95th
CAP percentil
PHC

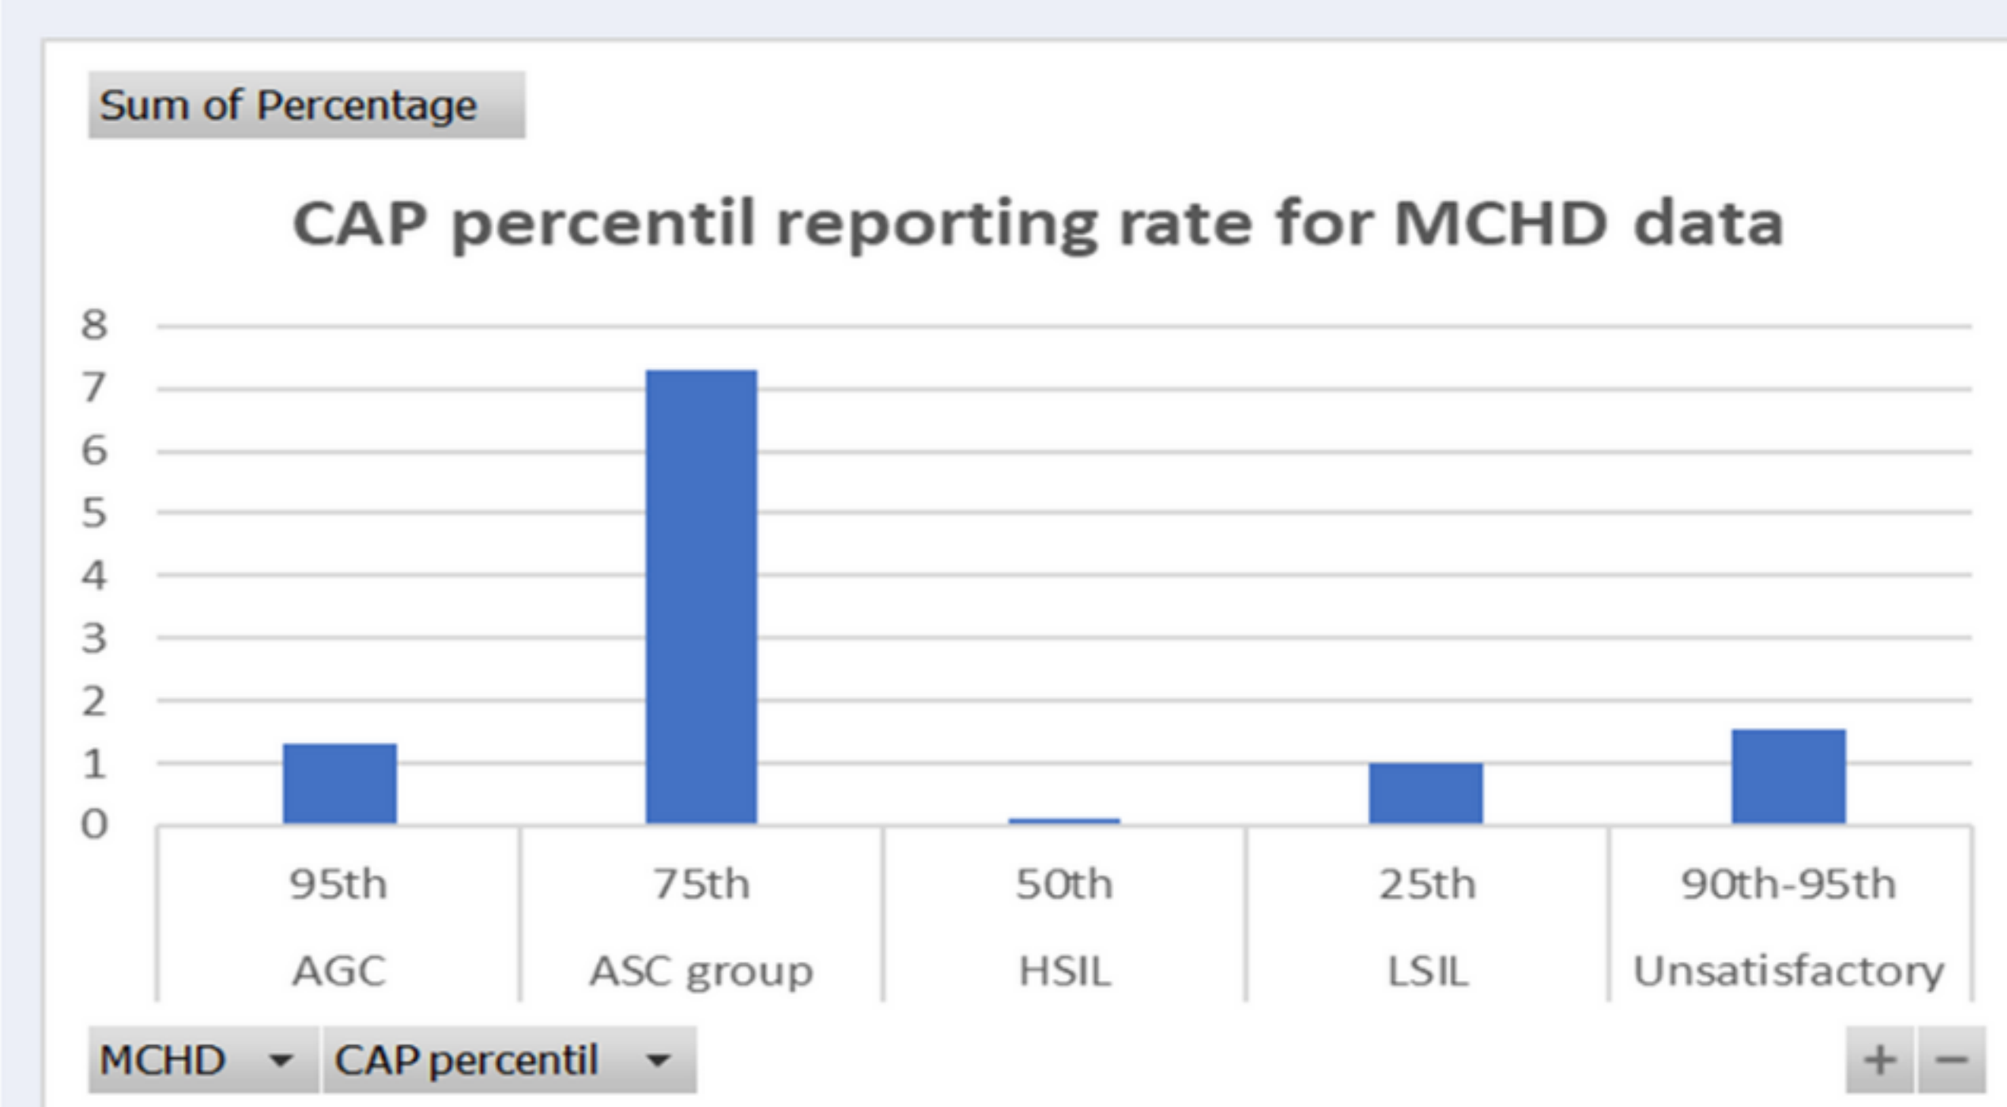

Sum of Percentage
CAP percentil reporting rate for MCHD data
95th
AGC
75th
ASC group
50th
HSIL
25th
LSIL
90th-95th
Unsatisfactory
MCHD
CAP percentil

Fig.4: Percentage of patients with positive cervical cancer results, and cervical cytology classification rates based on CAP classification rate percentile as a benchmark.

Cont.

By tracking these KPIs over time, program managers an evaluate the effectiveness of the cervical cancer screening program, identify areas for improvement, and make decisions to optimize early detection, treatment modalities, and prevention of cervical cancer

Conclusion: Recommendation: Documenting challenges and lessons at each phase of a cervical cancer screening program is crucial for refining implementation approaches. Pilot testing helps uncover and anticipating challenges during the scale-up phase, such as supply chain management for test kits. User-friendly digital tracking systems, such as RedCAP. Continuous improvement of screening tests and tools based on evolving technologies. Partnering with secondary institutions to explore introducing emerging technologies like HPV DNA testing can be considered for future scale-up phases. Standardizing pathological reports and assessments across implementers helps achieve long-term centralized coordination and strengthens referral pathways from screening to treatment. Upgrading lab facilities by improving infrastructure and installing state-of-the-art equipment leads to faster and improved healthcare services. In conclusion, Pap smear-based cervical cancer screening programs have the potential to reduce the burden of cervical cancer . However, successful implementation requires collaborative efforts between governments, healthcare organizations, and international partners are essential for developing sustainable and comprehensive cervical cancer screening programs that can make a significant impact on women's health in early staged cervical cancer through national health screening program in Saudi Arabia.

@E1_MCH