Digital Multidisciplinary Meetings show tangible benefits over live tumour boards

Primary: Carol-Ann Benn, MB.BCh (Wits) FCS (SA) (1)(2)
Second Author: Dr Charl J. van Loggerenberg, MB.BCh (Wits) FCEM (SA) MBA (Wits) Dip.PEC (SA) Dip.BM (DMS) (2)(3)
Assisting: Dominic Van Loggerenberg(1)
Assisting: Tanya Volschenk (1)
(1)Netcare Breast Care Centre of Excellence, Milpark Hospital, 9 Guild Road South Africa; (2)Faculty of Health Sciences, University of the Witwatersrand, Johannesburg South Africa. (3) Life Healthcare Group South Africa

BREAST CARE CENTRE
A Specialised Unit

NAPBC
NATIONAL ACCREDITATION PROGRAM FOR BREAST CENTERS
ACCREDITED BREAST CENTER
A QUALITY PROGRAM of the AMERICAN COLLEGE OF SURGEONS

Background

The Netcare Breast Care Centre has been operational and running as a multi-disciplinary breast care centre since 2000 and has been fully NAPBC accredited since 2016. The Covid19 pandemic necessitated changes in standard operating procedures, one of which was the conversion of the weekly tumour board meeting into a fully digital MultiDisciplinary Meeting (MDM), which resulted in a number of quality improvements leading to the decision to conduct virtual meetings permanently.

Methodology

After a rapid conversion period in early 2021, a dedicated system was established, allowing for accurate digital record-keeping, coupled with full recording and a digital clerking system. This created an environment that allowed for the meeting to host an increased number of members as certain logistical limitations were removed.

The virtual meeting was run in conjunction with representatives from the radiological and pathological members to allow for the presentation of all case-relevant radiology and pathology to be coupled with the recorded meeting and to be time-stamped, creating a complete reviewable database of all patients seen over the previous 18 months. The system thus also improved the accuracy of result collation.

Results

The meeting attendance rates averaged 70% for main members and 50% for secondary members; this rate equalised under the virtual MDM to over 85% for all members. This, coupled with being able to increase the scope and size of the meeting, allowed for over 30 newly diagnosed patients per week, over 15 Oncotype results and over 40 mid-treatment patients to be discussed weekly with 32 specialists.

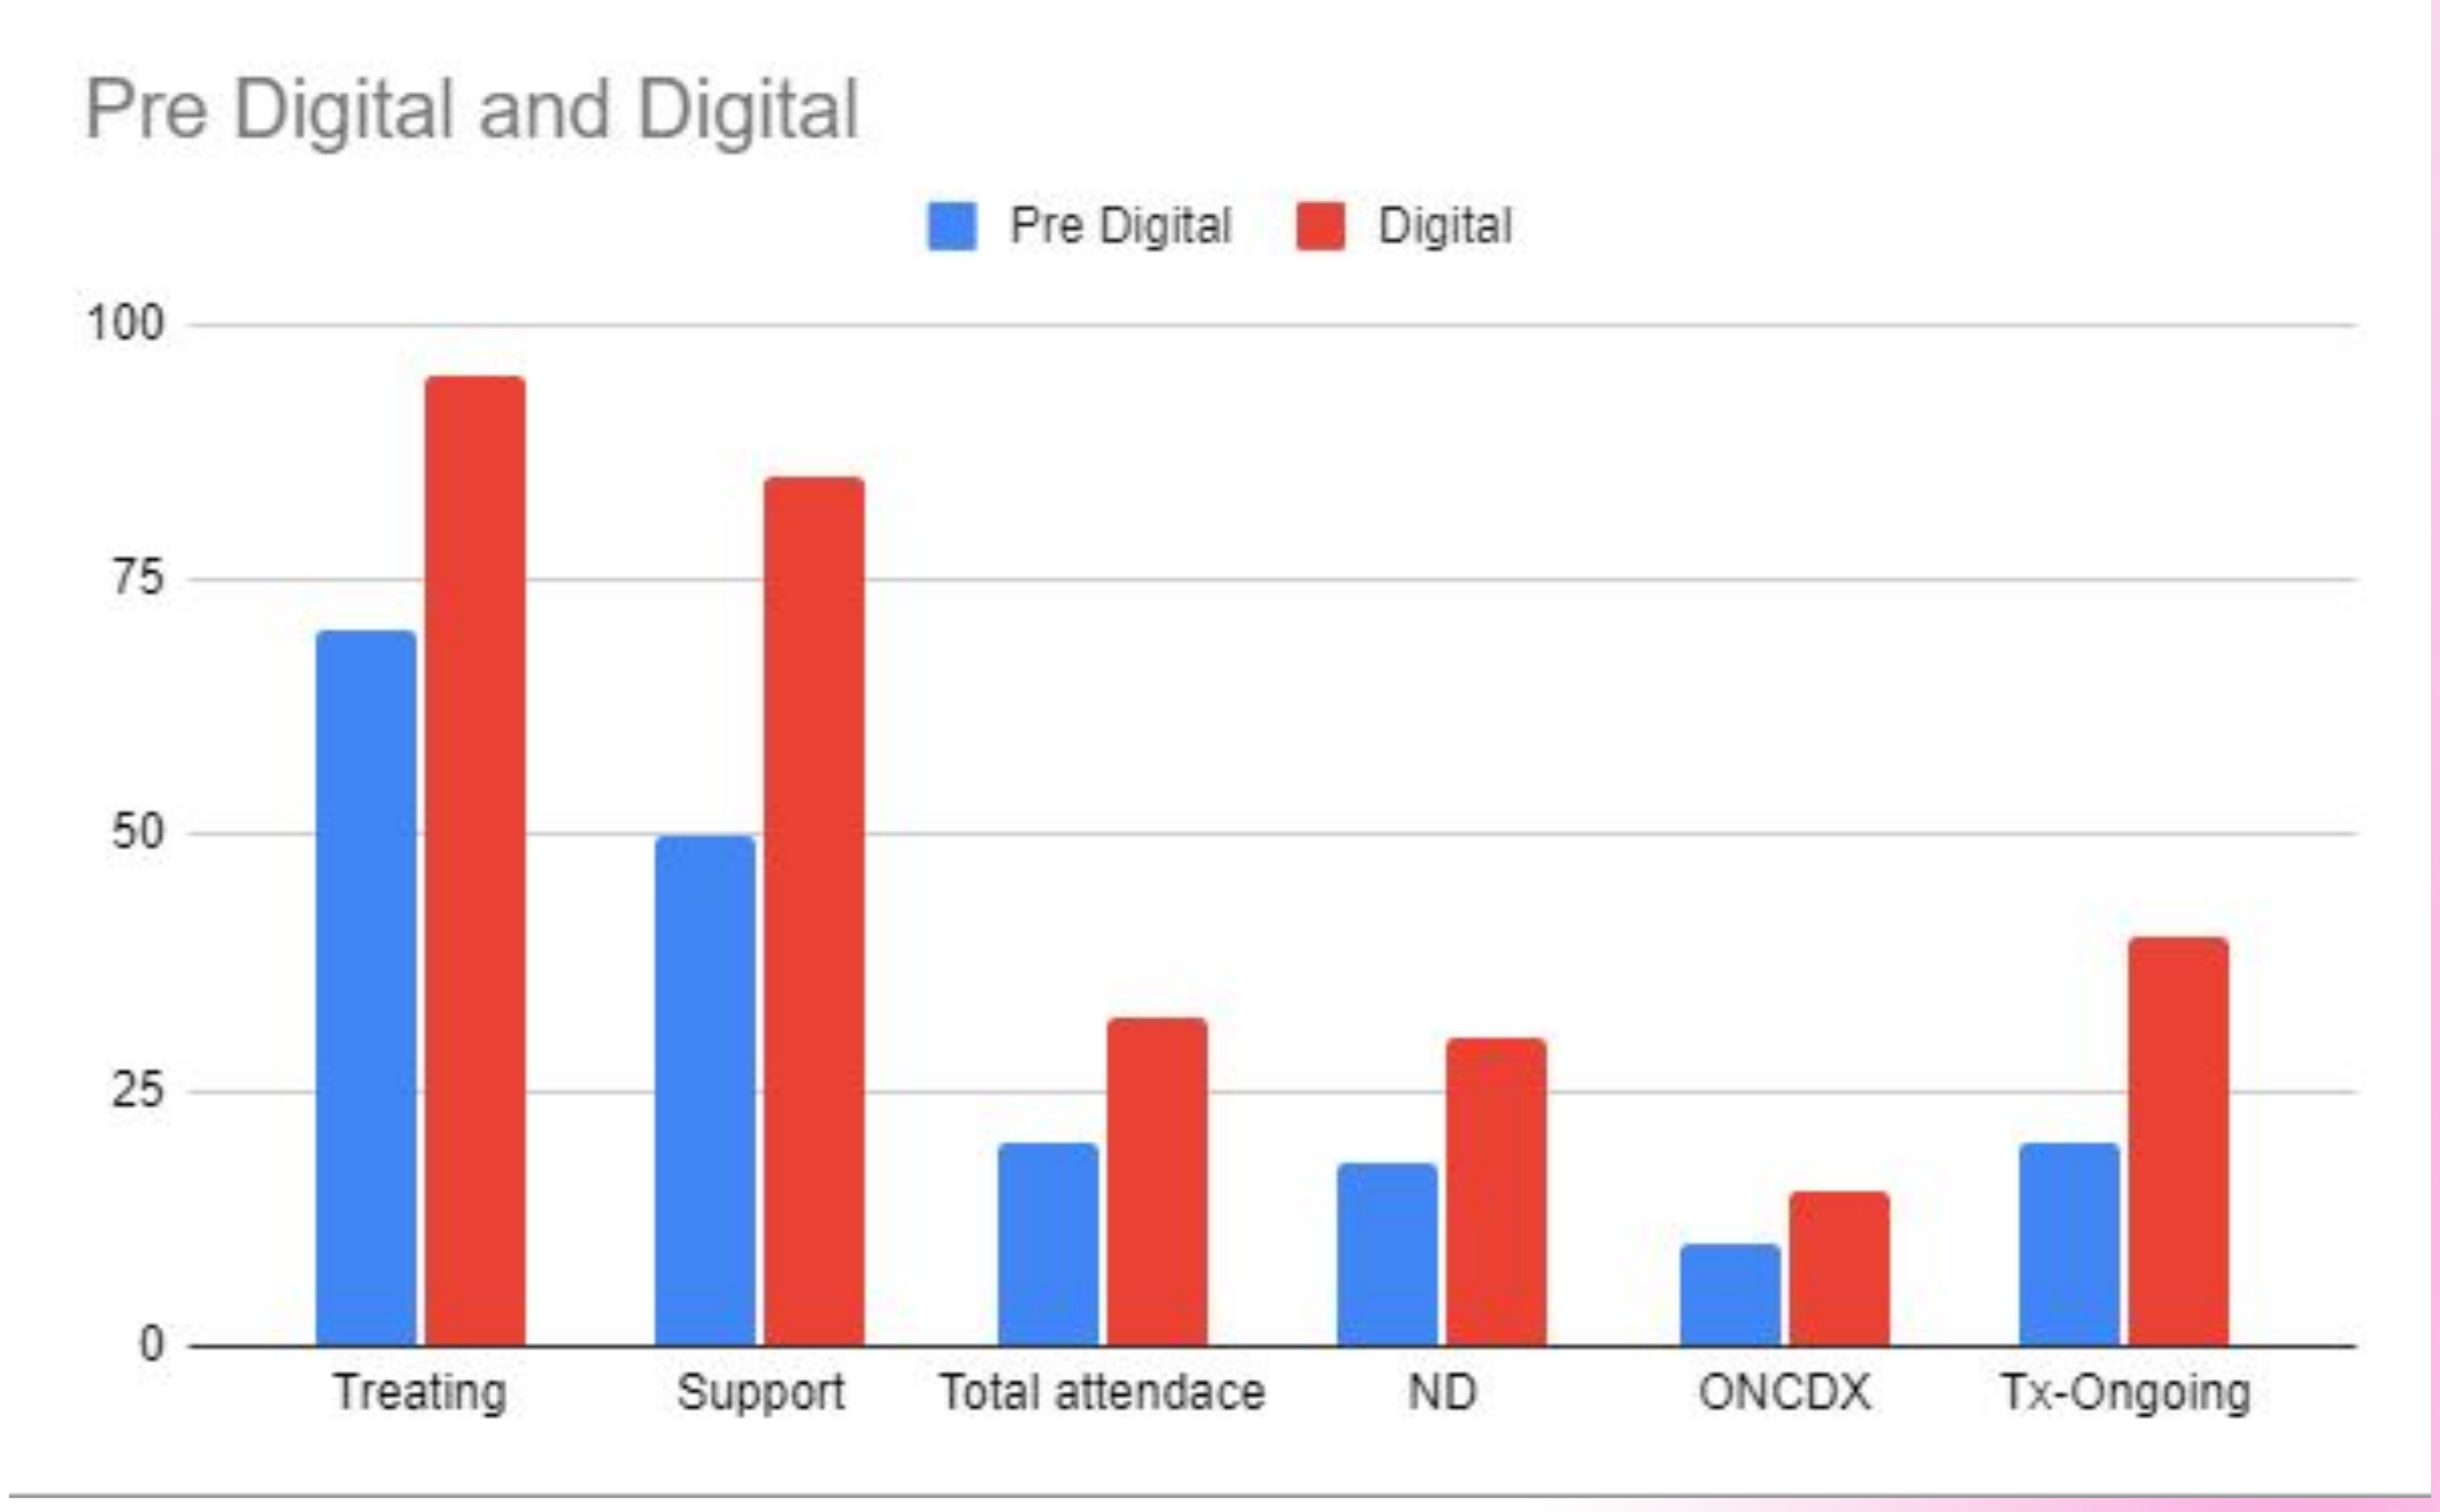
Pre Digital and Digital
Pre Digital
Digital
100
75
50
25
0
Treating
Support
Total attendace
ND
ONCDX
Tx-Ongoing

Conclusions

The Digital MDM environment has been unanimously chosen as the preferred option for the centre, with increased patient coverage and specialist access as clear benefits. Furthermore, the comprehensive record-keeping allows for the review of any patients seen for both treatment review and future-proofing any treatment decisions from a medicolegal standpoint.

ISQUA2023-ABS-3087