Kirsten Carter | kirsten.carter@novartis.com

Patient insights research exploring disease awareness, patient life experience, and current management of visceral leishmaniasis in Bihar, India

Kirsten Carter,[1] Shyam Sundar,[2] Koert Ritmeijer,[3] Margriet den Boer,[3] Clare Zamble,[4] Colin Forsyth,[5] Fabiana Alves,[6] Gerhild Angyalosi[1]

[1]Novartis Pharma AG, Basel, Switzerland; [2]Institute of Medical Sciences, Banaras Hindu University, Varanasi, India; [3]Médecins Sans Frontières, Amsterdam, Netherlands; [4]Lumanity, London, UK; [5]Drugs for Neglected Diseases initiative, New York, NY, USA; [6]Drugs for Neglected Diseases initiative, Geneva, Switzerland.

CONCLUSIONS

- Albeit in a relatively small sample size, this study reveals the barriers to disease awareness and access to care for patients with VL in the Bihar area.
- It shows that, while patients with VL seek care for symptoms early on, diagnosis follows, on average, 14 days following symptom onset.
- The current standard-of-care therapy is often not available in the healthcare center of diagnosis, which can further the delay in treatment administration.
- Oral therapy as an alternative to current parenteral treatment is preferred both by patients and caregivers of pediatric patients.

The study was funded by Novartis.

Poster presented at the European Congress on Tropical Medicine and International Health (ECTMIH) Biannual Meeting, November 20-23 2023, Netherlands, Amsterdam.

BACKGROUND

- Visceral leishmaniasis (VL) is mostly prevalent in rural areas and affects, almost exclusively, low-income populations with limited healthcare access.[1]
- There are little data on patients' perspectives, their diagnosis and treatment experiences, and how these experiences influence their expectations about treatment options.
- In India, the current standard-of-care therapy consists of 10 mg/kg single-dose liposomal amphotericin B by intravenous infusion. While highly efficacious, infusions are challenging in resource-limited settings and require close monitoring. Hence, liposomal amphotericin B treatment is currently limited to tertiary centers (restricted to public hospitals).[2]

Research objectives

- To gain insights regarding patients' experiences of the disease, diagnosis, and treatment.
- To better understand current barriers to disease awareness and diagnosis and treatment access.
- To explore acceptability of an oral treatment formulation among adult patients and caregivers of pediatric patients.

METHODS

A qualitative research study was conducted, using moderator-facilitated discussion, with a predefined set of screening criteria and following a protocol-defined discussion guide.

Research location

- Ten districts across Bihar, India, that account for the majority of VL cases.

Participants

- Eighteen adult patients and 12 caregivers of children who reported being diagnosed and treated for VL within the last 12 months were included.

Participant identification and interviews

- District hospitals were visited to identify villages/townships of hotspots of patients with VL in past 24 months (per study eligibility criteria).
- Recruiters met with local pharmacists, community healthcare professionals (HCPs), and local leaders/village council members to confirm relevance of target areas.
- Recruitment was conducted door-to-door, where consent was gained for screening.
- Patients were recruited via a screening questionnaire to determine eligibility and willingness to take part in the interview.
- Self-reported VL of participants/caregivers: 12/30 patients/caregivers had a VL card; 13/30 had lost/no longer possessed a VL card; 5/30 were unable to confirm diagnosis.
- Sixty-minute, face-to-face, in-depth interviews were conducted in Hindi by a local partner agency.

RESULTS

Figure 1. Summary of life experiences of 18 adults and 12 caregivers of children with VL

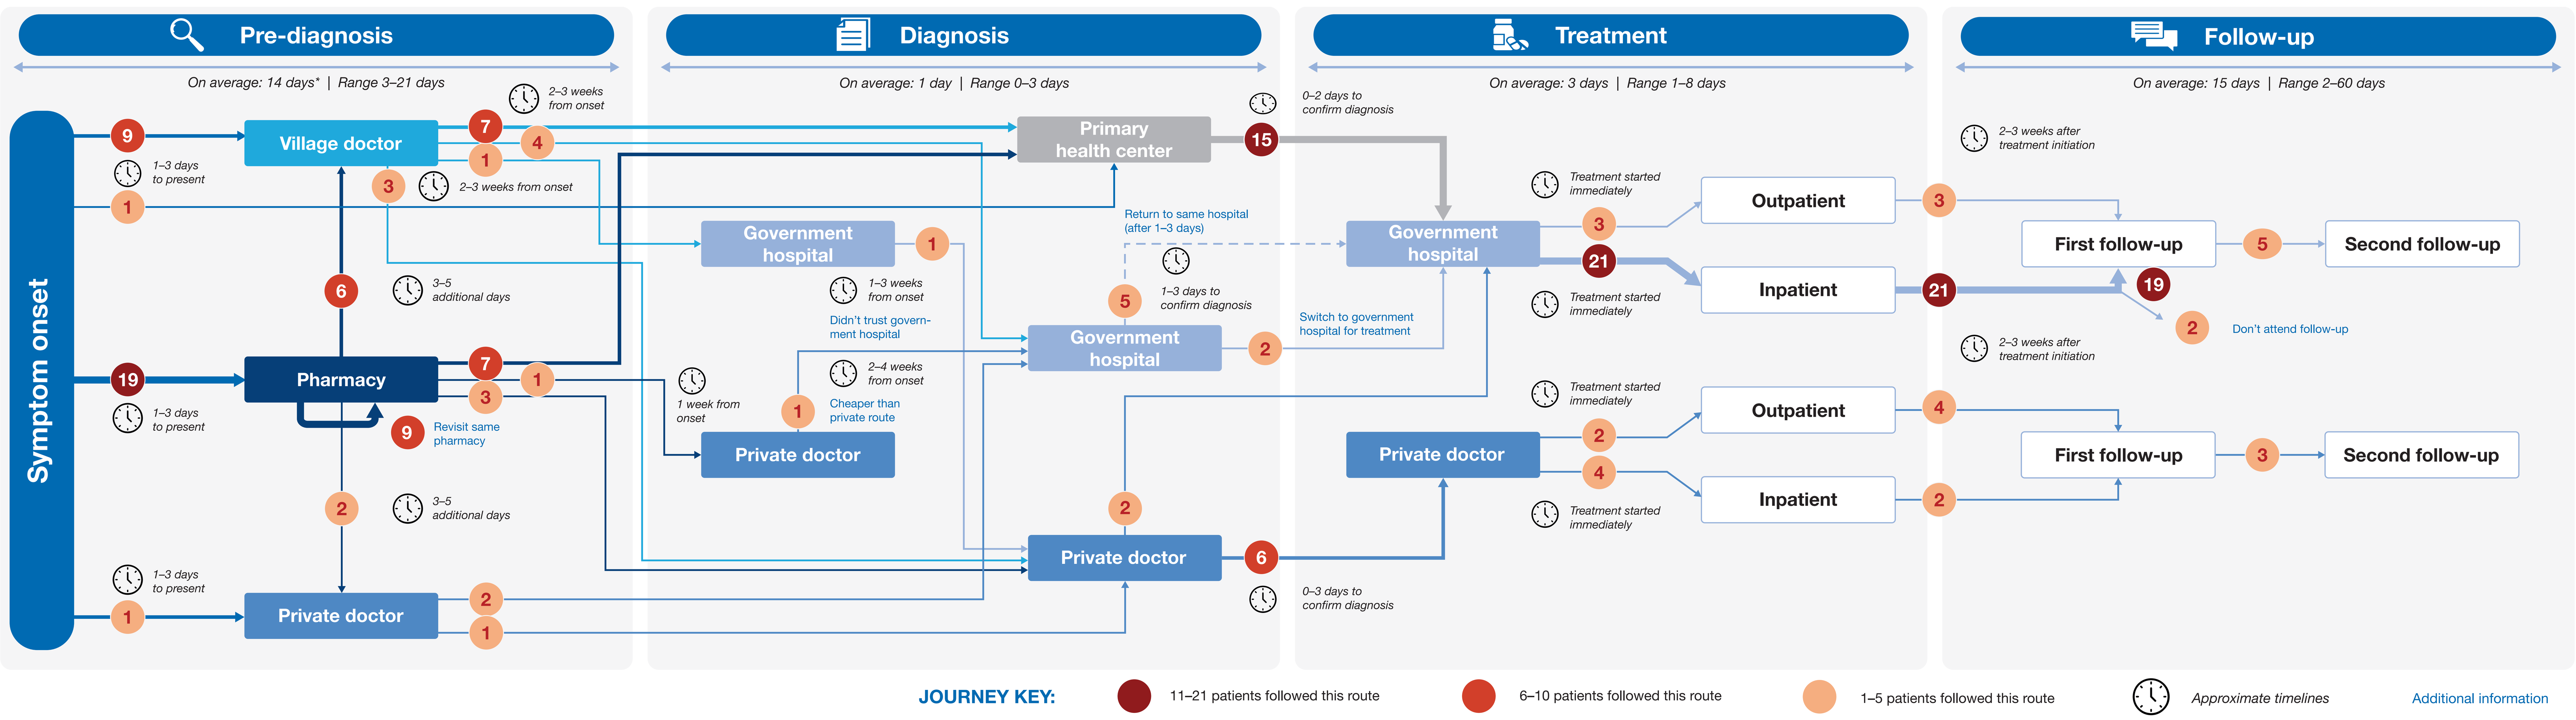
Pre-diagnosis
On average: 14 days* | Range 3–21 days
Diagnosis
On average: 1 day | Range 0–3 days
Treatment
On average: 3 days | Range 1–8 days
Follow-up
On average: 15 days | Range 2–60 days
Symptom onset
Village doctor
Pharmacy
Private doctor
1–3 days to present
2–3 weeks from onset
3–5 additional days
Revisit same pharmacy
Primary health center
Government hospital
Private doctor
1–3 weeks from onset
Didn't trust government hospital
2–4 weeks from onset
Cheaper than private route
1 week from onset
Return to same hospital (after 1–3 days)
1–3 days to confirm diagnosis
0–2 days to confirm diagnosis
0–3 days to confirm diagnosis
Switch to government hospital for treatment
Treatment started immediately
Outpatient
Inpatient
2–3 weeks after treatment initiation
First follow-up
Second follow-up
Don't attend follow-up
JOURNEY KEY:
11–21 patients followed this route
6–10 patients followed this route
1–5 patients followed this route
Approximate timelines
Additional information

*Excluding one anomaly of 90 days. Mean, median, and range breakdowns (with and without outlier) below.
- Mean 16.4 days (with outlier); 13.8 days (without outlier).
- Median 12 days (with outlier); 7.5 days (without outlier).
- Range 3–90 days (with outlier); 3–21 days (without outlier).
VL, visceral leishmaniasis.

Figure 2. Breakdown of patients' experiences of VL

	Onset of symptoms	First touchpoint(s) – primary care	Present to diagnosing clinic	Receive diagnosis and treatment	Treatment initiation	Symptoms resolved	Follow-up
Timeline	Day 0	First touchpoint within **3 days*** of experiencing symptoms (range 1–25 days)	**~2–3 weeks** after first experiencing symptoms (range 2–100 days)	**+1 day** on average (range 0–3 days)	0–2 days on average	Patients started feeling better ~1 week after treatment was initiated (range 1 day–2 months)	All patients offered follow-up, at variable periods (range 2 days–2 months)
Additional detail	Symptoms include fever, body pain, fatigue, loss of appetite	Patients seek advice from local medical shop/pharmacist and given oral treatment – i.e., paracetamol Half then visit another primary care setting (i.e., primary health centers or village doctor) after a couple of days of no improvement Some patients (n=9) revisit the same pharmacy multiple times	n=26 specify receiving a blood test for diagnosis and n=7 also describe a clinical assessment and/or urine sample Happens across a range of clinics (n=15 primary health centers, n=7 government hospitals, n=8 private) In government setting, patients often prescribed treatment for symptoms (non-specific to VL) while waiting for results	Small delay of several days to confirm diagnosis while diagnostic tests are run, in both private and government hospital settings	65% switch to a different practice setting for treatment (n=19) and therefore experience a delay of 1–2 days in starting treatment. Otherwise, treatment initiated immediately	~1 week after treatment is completed, patients start to resume normal activities/work	Majority of patients (n=18) attend one follow-up. Eight patients report attending a second follow-up Consultation consists of a blood test and potential treatment for strength

*Median 3 days; mean 3.8 days.
VL, visceral leishmaniasis.

Key learnings on patients' life experiences

Pre-diagnosis

- Low awareness indicates VL is not suspected at early stages of disease (as potentially other infectious conditions are being considered).
- Low sense of urgency at first point of care level (usually pharmacists or village doctors) delays referral to a hospital/diagnostic setting (**Figure 1**).

Diagnosis

- Half of the patients received their diagnosis at a primary or community healthcare center, where treatment for VL is not available. Delay in treatment administration can be up to 2 days as a result of the need to switch treatment facility (**Figure 1**).
- The time from symptom onset to diagnosis (2–3 weeks in most cases) is in line with the Indian national program recommendation.

Treatment and follow-up

- While treatment is free in a government setting, a few patients (n=6) choose to have treatment in a private healthcare setting (**Figure 1**).
- Some stigma or negativity is associated with going to government facilities, in terms of hygiene, quality of care, and waiting times.
- The majority of patients (n=27) described receiving infusion treatment, but patient descriptions indicate single-dose liposomal amphotericin B may not have been used in all cases (**Figure 2**).
- Post-treatment, the majority of patients (n=28) attend one follow-up appointment and understand their VL to be resolved.

Potential biases and study limitations

- The diagnosis of VL is based on self-reporting. The majority of respondents were not able to recall specific treatment details (frequency and duration); patients were unable to distinguish VL-specific treatment from supportive treatment.
- Findings may not be reflective of other urban populations or populations from other parts of India.
- The caregiver sample reflects only the experience of caregivers of pediatric patients aged 10–14 years.

Learnings regarding a potential new oral treatment

There is an unmet need for new oral, efficacious, safe, well-tolerated, convenient, easy-to-use drugs in VL, and, currently, research is ongoing to identify and develop new drugs addressing this need in patients with VL.

Adult participants' feedback

- Most respondents would choose oral therapy over that received, but some perceive that the tablets would have a slower onset of action and it may take longer to feel better/be cured.
- Both treatment durations discussed (7 days and 14 days) were acceptable, with an overall preference for 7 days.
- Participants may have limited finances. A few respondents stated that they would be willing to pay a maximum of $72 (₹5985) from their pocket for their treatment course.

Caregivers' feedback

- Good acceptability of a hypothetical pediatric treatment (capsule containing mini-tablets, which could be added to a spoonful of a child's food): most think mini-tablets would work for their children.
- Oral route preferred vs injections/infusions by most caregivers (11/12), to avoid painful injections.
- Oral therapy preferred as it would presumably not require HCP administration or travel.
- Caregivers mentioned they would pay up to $60 (₹4987) for this treatment.

Acknowledgments

- Thanks to all of the participants of this research, and to Kadence for conducting the interviews in Bihar.
- Bethan Hughes, Shaishav Panchal, and Andreas Tietz of Novartis, and Maddie McLeod and Fleur Bennetts of Lumanity, are all acknowledged for their efforts supporting the design, review, execution, and analysis of this research.
- All authors participated in the development of this poster and approved the final poster for presentation.

Disclosures

K Carter is an employee of Novartis Pharma AG and received stock options from Novartis Pharma AG. **S Sundar** has received consulting fees from Novartis Pharma AG. **K Ritmeijer** has no conflict of interest to disclose. **M den Boer** has no conflict of interest to disclose. **C Zamble**'s company (Lumanity) has been contracted and paid by Novartis Pharma AG for writing support and market research. **C Forsyth** is employed by Drugs for Neglected Diseases initiative (non-profit organisation). **F Alves** is employed by Drugs for Neglected Diseases initiative (non-profit organisation). **G Angyalosi** is an employee of Novartis Pharma AG and received stock options from Novartis Pharma AG.

References

1. Sundar S, *et al. Expert Rev Anti Infect Ther* 2018;16:805–812.
2. Singh OP, *et al. Infect Dis Poverty* 2016;5:19.