Interpreting Clinical Meaningfulness of SP-102 for the Treatment of Lumbosacral Radicular Pain (LRP): A Post-hoc Analysis of the CLEAR-1 Trial and A Systematic Review of Literature

Miller AM, Knezevic NN, Nalamachu S, Meske D, Ambrose C, Vought K, Chan EK, Lissin D

BACKGROUND

Analgesic clinical studies performed for the purposes of FDA approval produce results that require interpretation to fully understand their clinical meaningfulness. The results of these studies are often misinterpreted with the most common source of confusion arising when the magnitude of group differences are conflated with the determination of the magnitude of improvement within subjects that can be considered clinically important[1,2,3]. While there has been consensus on what constitutes a clinically meaningful change with-in individual subjects, group differences should include evaluations based on the overall risk-benefit profile of the treatment within the context of other available treatments for the indication[4].

Lumbosacral radicular pain (LRP), is a common and debilitating disorder that affects millions of Americans in their lifetime (13 to 40%)[5], leading to short- and long-term disability and increased healthcare costs[5,6]. There are no products approved by FDA to treat LRP. When conservative methods fail, patients are often referred for epidural steroid injections (ESIs) as a last line before surgery[7]. ESIs are used off-label in the absence of any Class I data and are associated with significant risks, including neurological complications and death, as outlined in the product labels.

SP-102 (10 mg of dexamethasone sodium phosphate in 2 mL of viscous gel solution) is an investigational non-particulate, preservative-free injectable product designed to increase the residence time of dexamethasone at the site of injection without increasing systemic exposure[8]. Recently, the CLEAR Trial (Corticosteroid Lumbosacral Epidural Analgesia for Radiculopathy) investigated the safety and efficacy of single and repeat SP-102 transforaminal injection (TFI) compared to a placebo sham intramuscular (IM) injection in subjects with LRP[8].

OBJECTIVE

Interpret the clinical meaningfulness of the magnitude of group mean differences (GMD) of the CLEAR-1 Trial by comparing to the effects observed for other analgesic products and in the context of overall safety.

METHODS

CLEAR Trial Population and Procedures:

- The CLEAR Trial was a double-blind, randomized, placebo-controlled, multicenter study conducted in the US (January 2018 - January 2022). The primary results were previously reported[8]. Subjects were screened for current episodes of LRP and randomized (1:1) to receive either a TFI of SP-102 (active) or an intramuscular (IM) placebo injection and followed for 24 weeks. Fluoroscopic guidance and verification of epidural contrast spread was used to confirm study drug administration. Subjects that did not have verified needle placement and contrast spread were excluded from the modified intent-to-treat (mITT) analyses (pre-defined in the statistical analysis plan (SAP). The intent-to-treat (ITT) population included all randomized subjects.

CLEAR Trial Endpoints:

- The primary endpoint was change in average daily pain on the Numeric Pain Rating Scale (NPRS) in the affected leg over 4 weeks. This publication also includes results from other pre-defined and post-hoc endpoints: (1) responder analysis, (2) Oswestry Disability Index (ODI) group mean comparisons, and (3) Brief Pain Inventory (BPI) Pain Interference group mean comparisons. The ODI and BPI clinically meaningful cutoff comparisons are post-hoc, the remainder were pre-defined.

Systematic Reviews:

Comparison to Other Approved Analgesics

- Because there are no approved products for LRP, Phase 3 efficacy results for existing analgesic products used to treat chronic low back pain (CLBP) were collected through publication and regulatory documents for the group mean difference (GMD) and standardized effect size (SES; Cohen's D). Pre-defined criteria were used to select a representative single result to present when multiple studies or doses were available based on common study design, highest approved dose, highest or middle result for the relevant dose.

Comparison to Other Transforaminal Epidural Steroid Injections

- Primary efficacy results (GMD and SES) from a recent Cochrane review that examined off-label ESIs, were collected for fluoroscopically verified transforaminal injections at 4 Weeks (or closet timepoint) and presented.

Statistical Analyses:

- The primary endpoint comparison used a linear contrast of the least square (LS) means comparing the weekly mean scores up to Week 4 (average of Weeks 1, 2, 3, and 4 change from baseline LS means estimated from the model). Responder analyses for NPRS Responder, ODI, and BPI Pain Inventory utilized chi square tests.
- Cohen's D was used for SES calculations and is determined by calculating the mean difference between the active and placebo groups (i.e., Group A mean – Group B mean) and then divided by the pooled standard deviation (SD).

RESULTS

The CLEAR Trial demonstrated a statistically significant difference between SP-102 and placebo for the primary endpoint and many secondaries for both populations (mITT and ITT) and supported the overall safety of the product[8] (Figure 1).

Clinically Meaningful With-in Subject Results:

- There are established consensus cutoffs for core CLBP analgesic clinical trial endpoints for with-in subject clinical meaningfulness, however the magnitude of between group differences (i.e., versus placebo) is not established (NPRS Responder Analyses [≥30% and ≥50% improvement], ODI [≥10-pt improvement], and BPI-PI [≥1-pt improvement]); results are presented to contextualize the with-in study results. These results establish that there were clinically meaningful improvements with-in subjects in a greater percentage of subjects in the SP-102 arm versus placebo, mostly statistically significant.

Figure 1. Percentage of participants with clinically meaningful reductions in relevant outcome measures

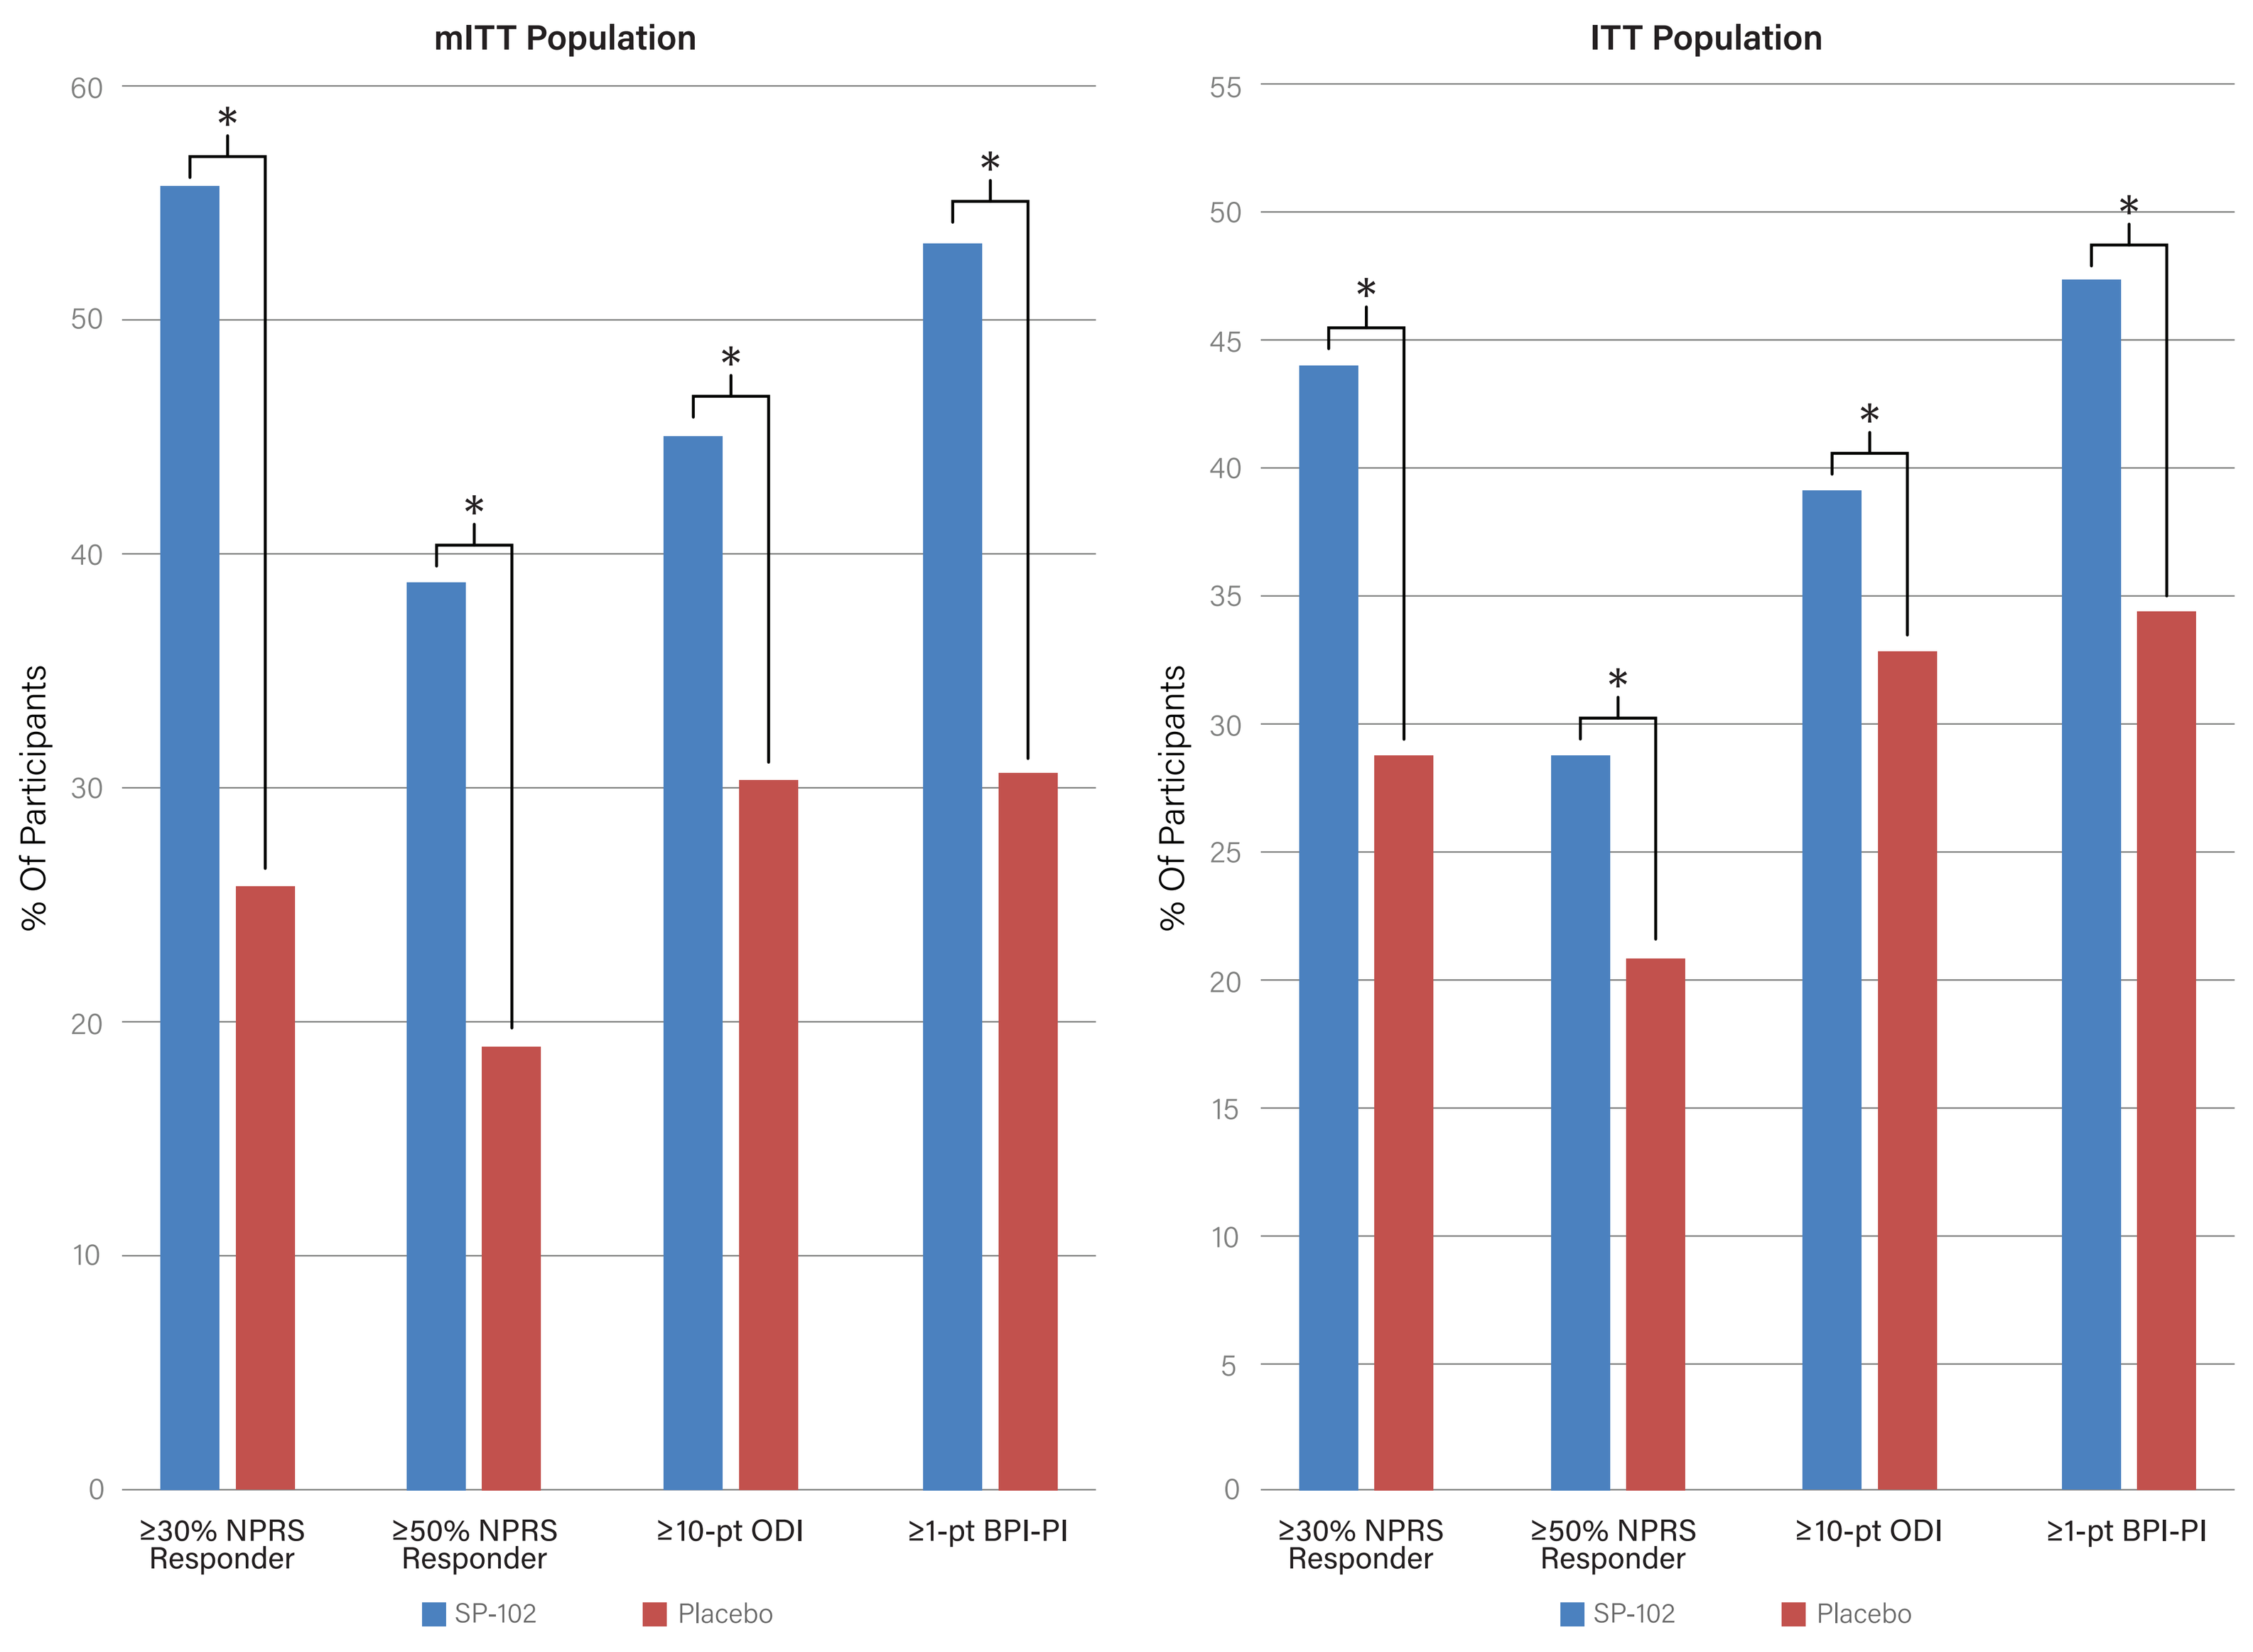
mITT Population
ITT Population
% Of Participants
≥30% NPRS Responder
≥50% NPRS Responder
≥10-pt ODI
≥1-pt BPI-PI
SP-102
Placebo
*

Abbreviations: BPI-PI = Brief Pain Inventory – Pain Intensity; NPRS = numerical pain rating scale (0-10); ODI = Oswestry Disability Index
*=p<0.05

Clinically Meaningful Between Group Differences:

Comparison of SESs for Currently Approved Analgesics:

- SES and NPRS GMD between active and placebo across products were compared (Figure 2a and 2b).

Figure 2a
Comparison of Standardized Effect Size In Chronic Low Back Pain Studies for the Primary Endpoint

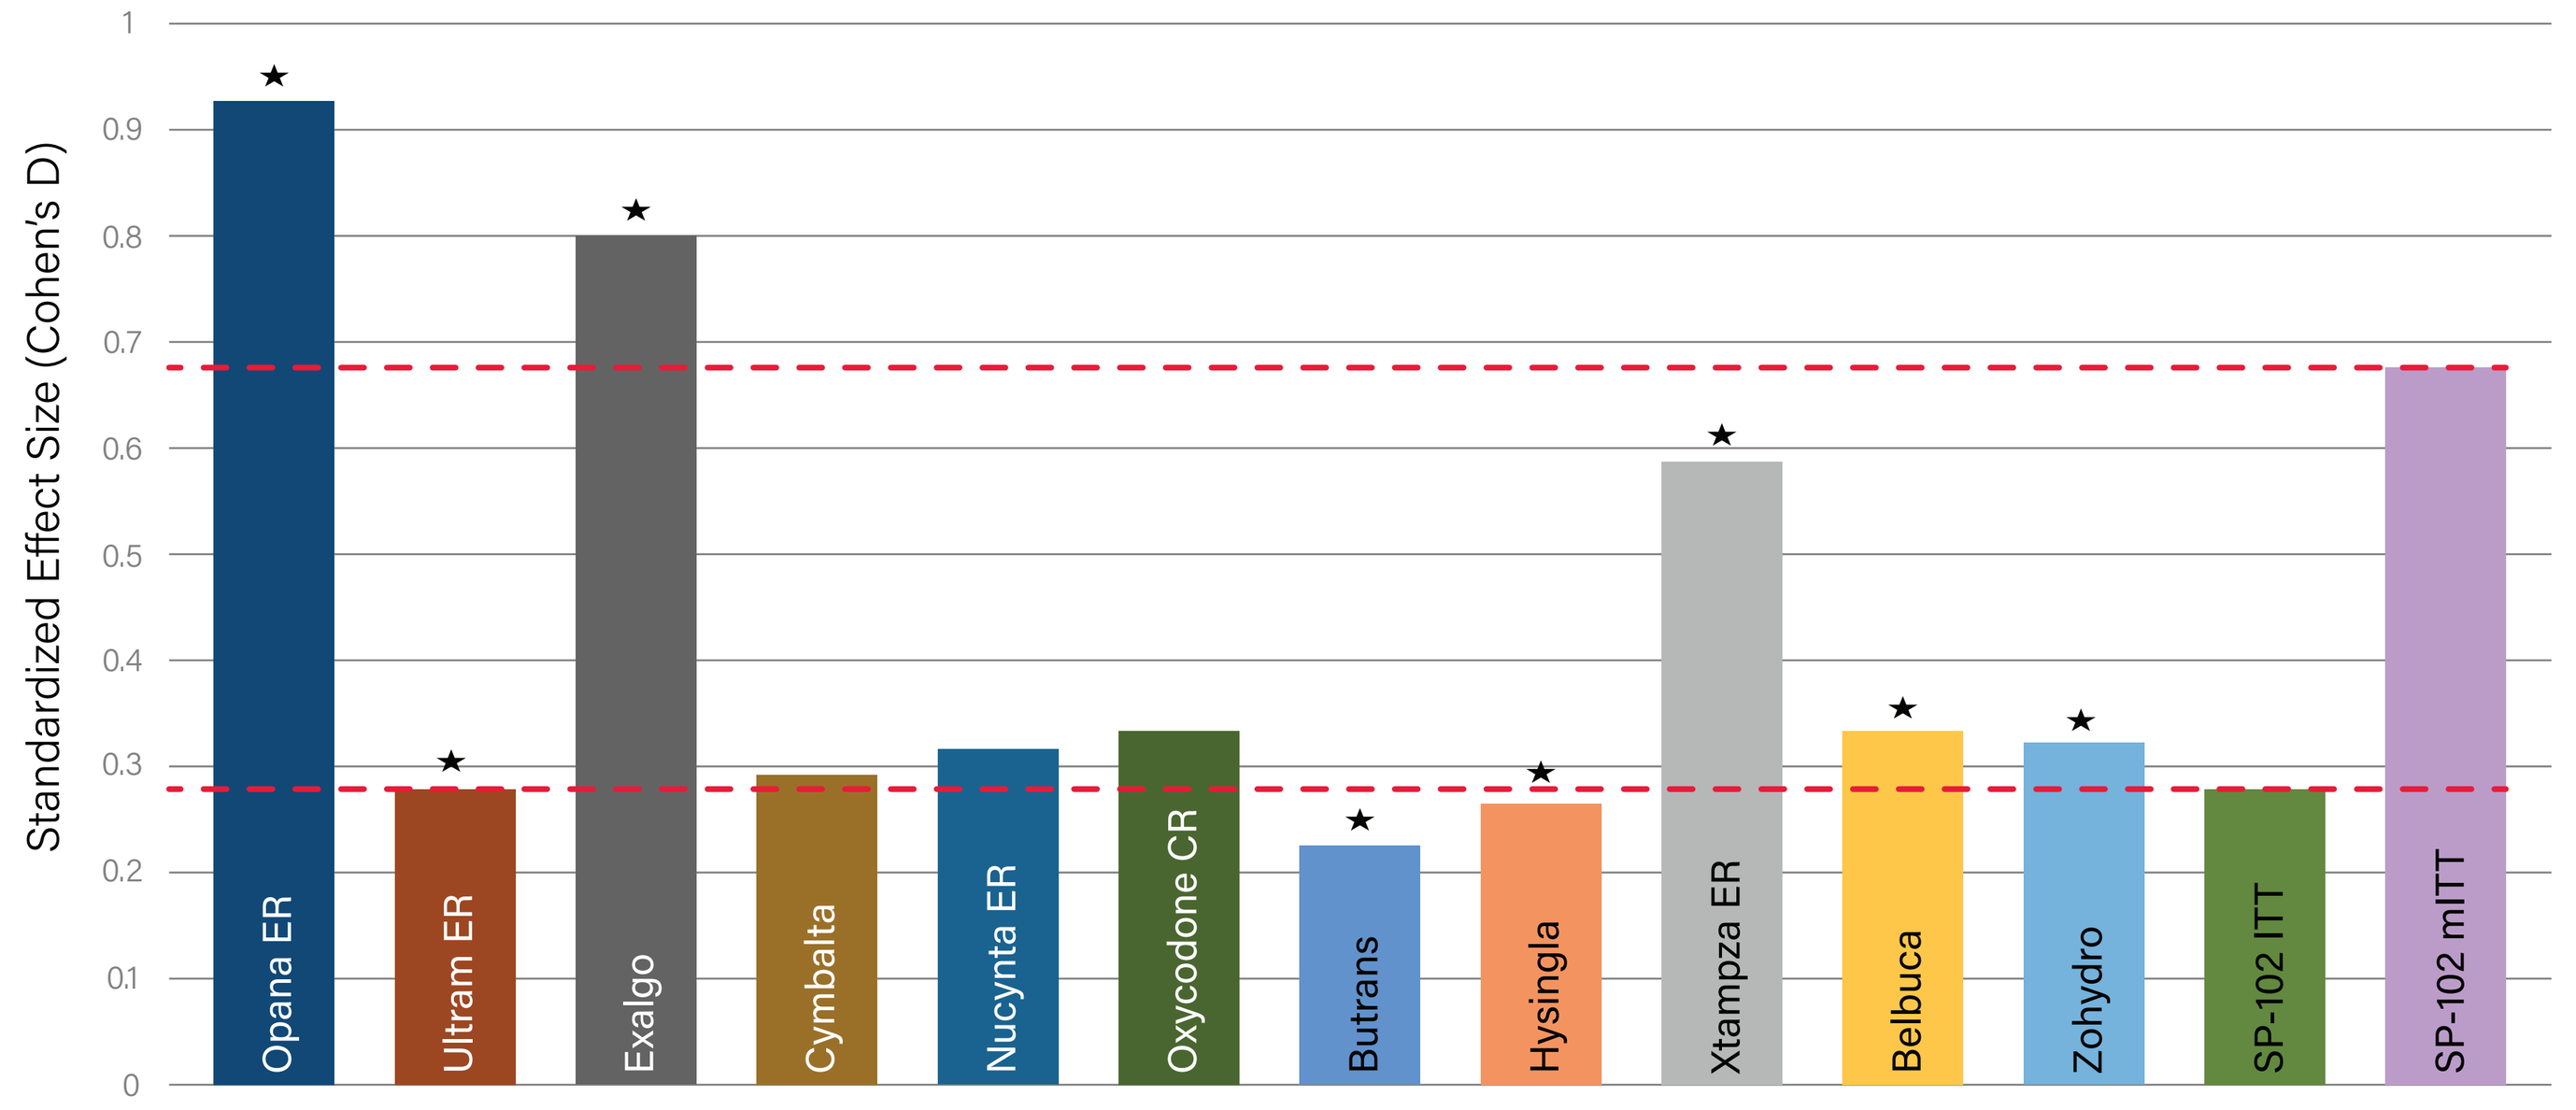
Standardized Effect Size (Cohen's D)
Opana ER
Ultram ER
Exalgo
Cymbalta
Nucynta ER
Oxycodone CR
Butrans
Hysingla
Xtampza ER
Belbuca
Zohydro
SP-102 ITT
SP-102 mITT

Figure 2b
Comparison of Between Mean Group Differences in Chronic Low Back Pain Studies on NPRS Scale at Primary Timepoint

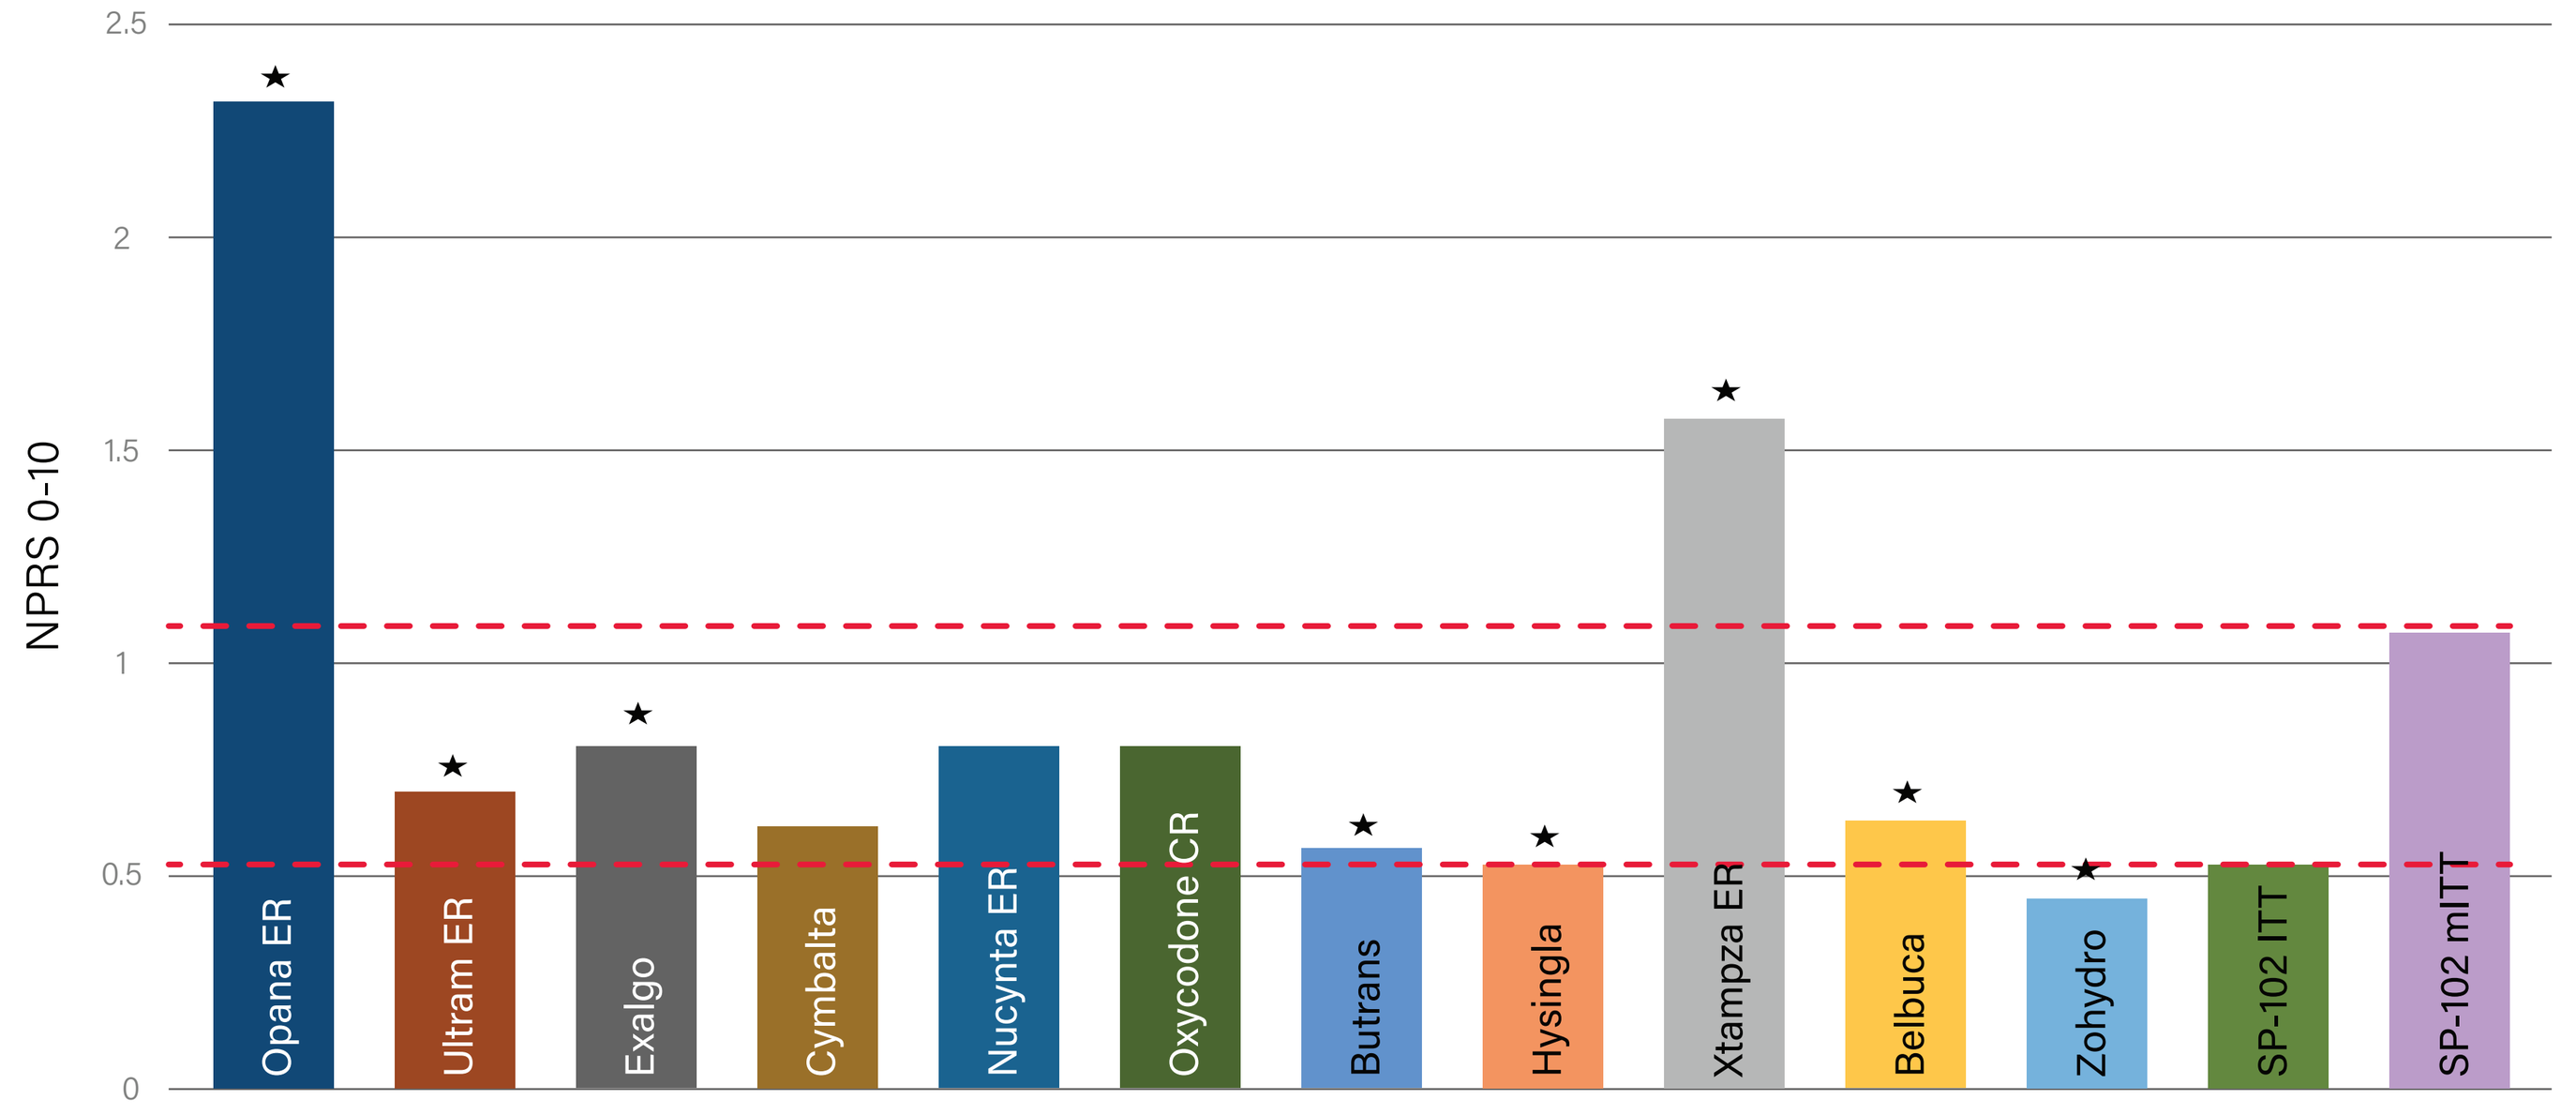
NPRS 0-10
Opana ER
Ultram ER
Exalgo
Cymbalta
Nucynta ER
Oxycodone CR
Butrans
Hysingla
Xtampza ER
Belbuca
Zohydro
SP-102 ITT
SP-102 mITT

Results for the mITT population (SES = 0.68) for SP-102 produced the 4th highest SES among currently approved analgesics (Figure 2A, mITT). In the ITT population, the SES was aligned with other products (SES = 0.28) (Figure 2A). Results were similar when NPRS GMD were compared, with results for the mITT population ranking 3rd (1.08-point difference).

Clinically Meaningful Between Group Differences:

Comparison of SESs for Studies of Off-Label Use of TFI Administered Steroids:

Figure 3a
Comparison of Standardized Effect Size

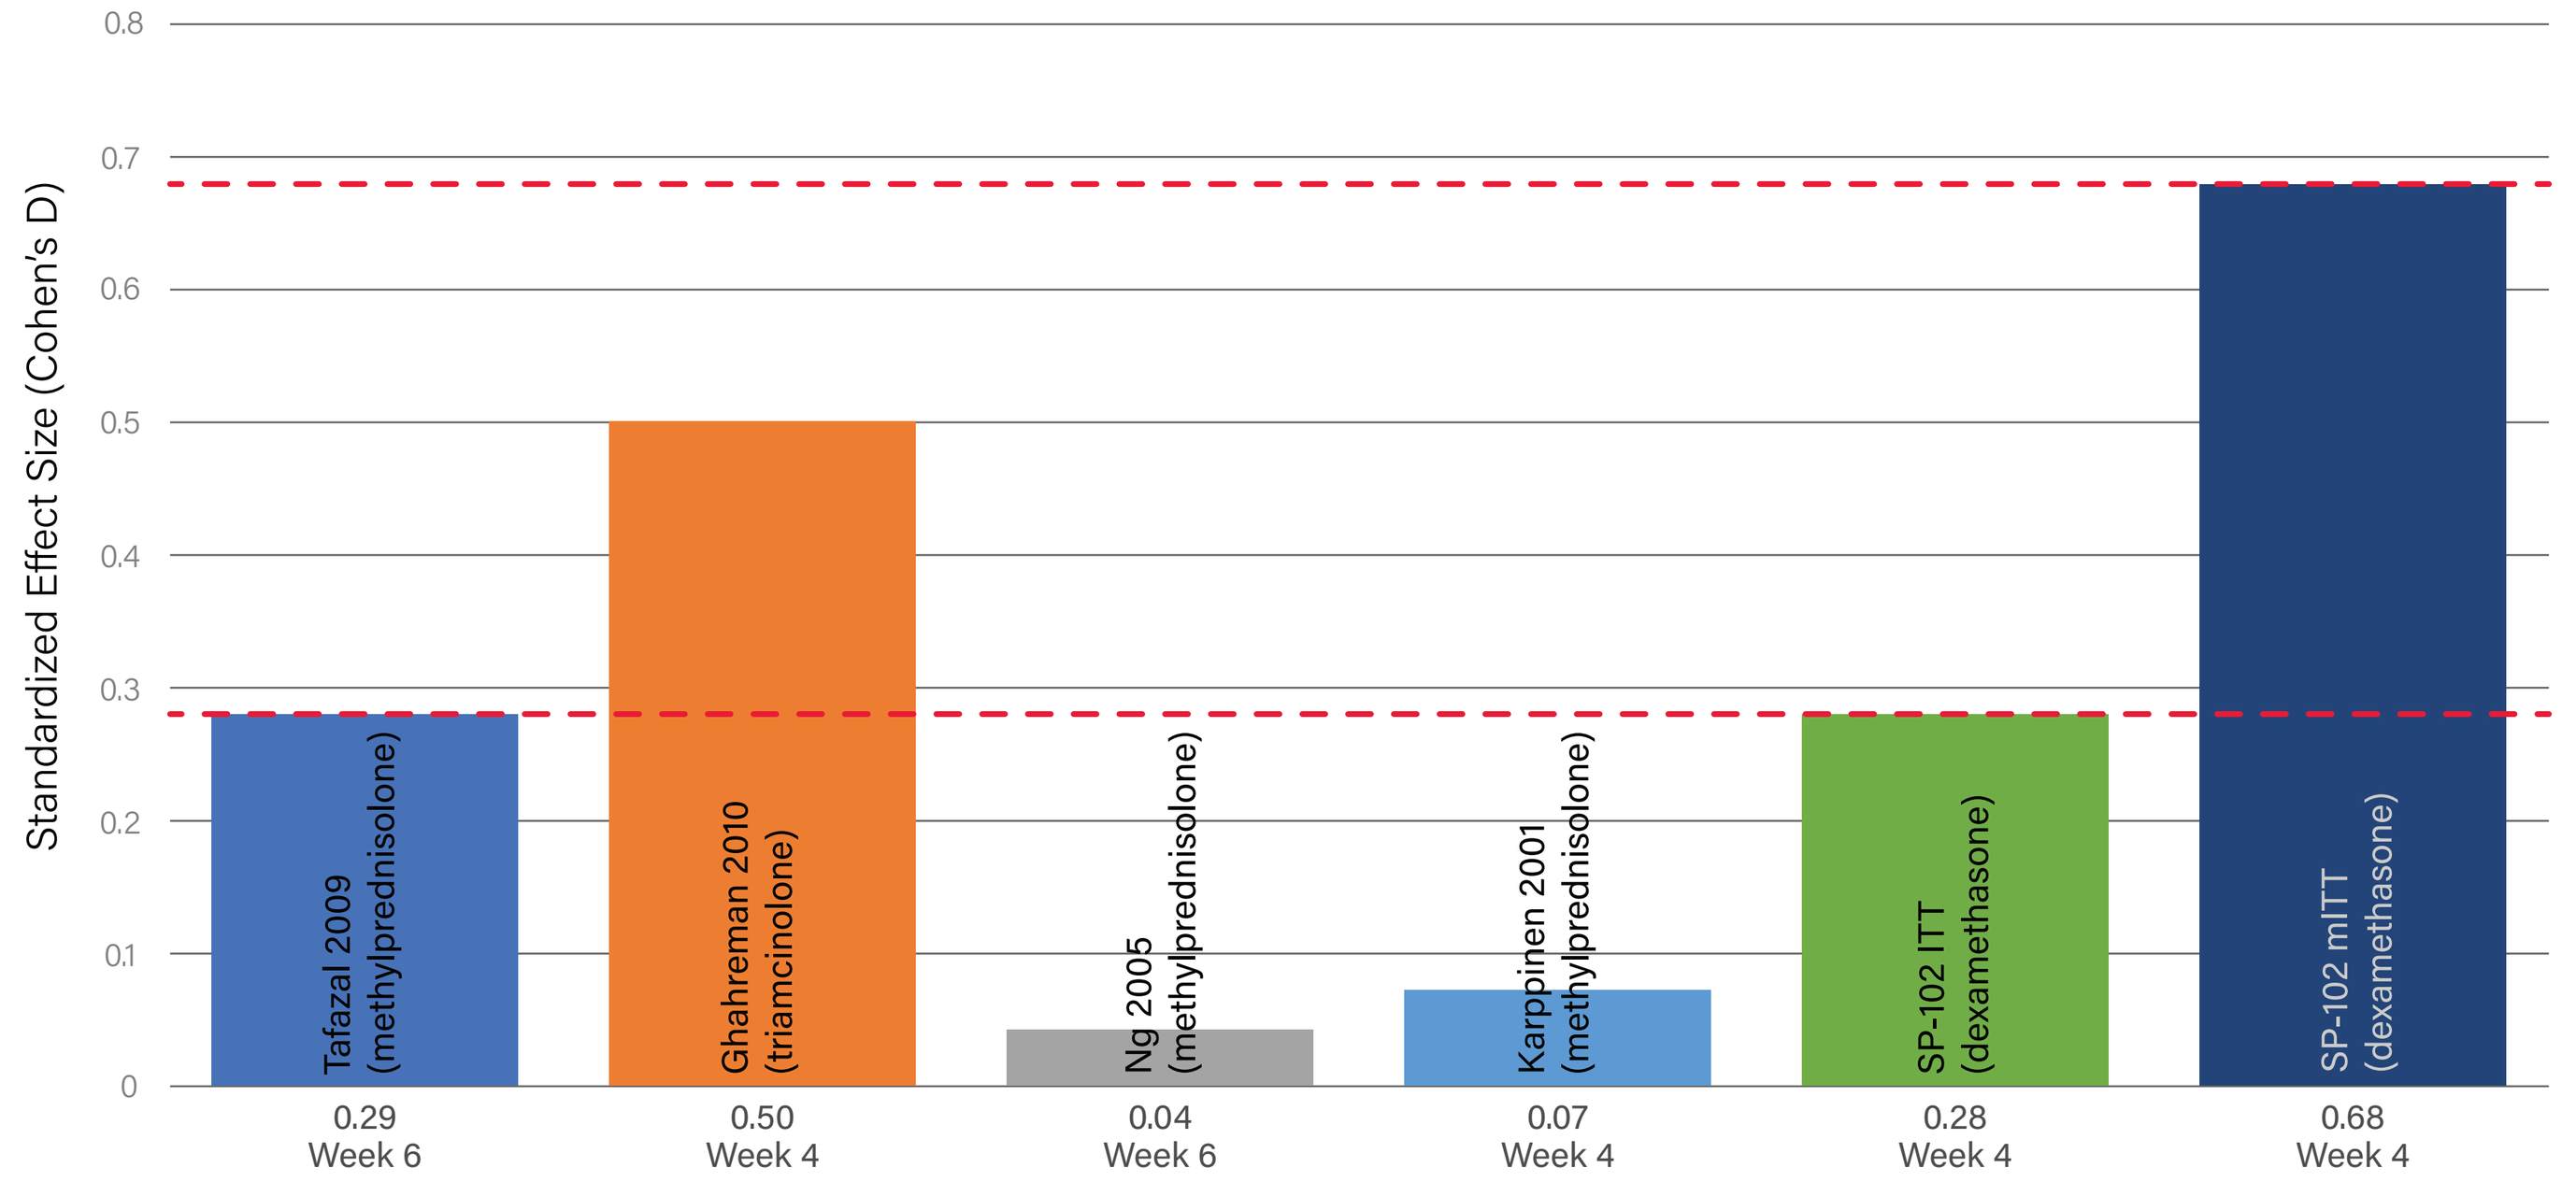
Standardized Effect Size (Cohen's D)
Tafazal 2009 (methylprednisolone)
0.29
Week 6
Ghahreman 2010 (triamcinolone)
0.50
Week 4
Ng 2005 (methylprednisolone)
0.04
Week 6
Karppinen 2001 (methylprednisolone)
0.07
Week 4
SP-102 ITT (dexamethasone)
0.28
Week 4
SP-102 mITT (dexamethasone)
0.68
Week 4

Figure 3b
Comparison of Between Mean Group Differences on NPRS Scale at Primary Timepoint

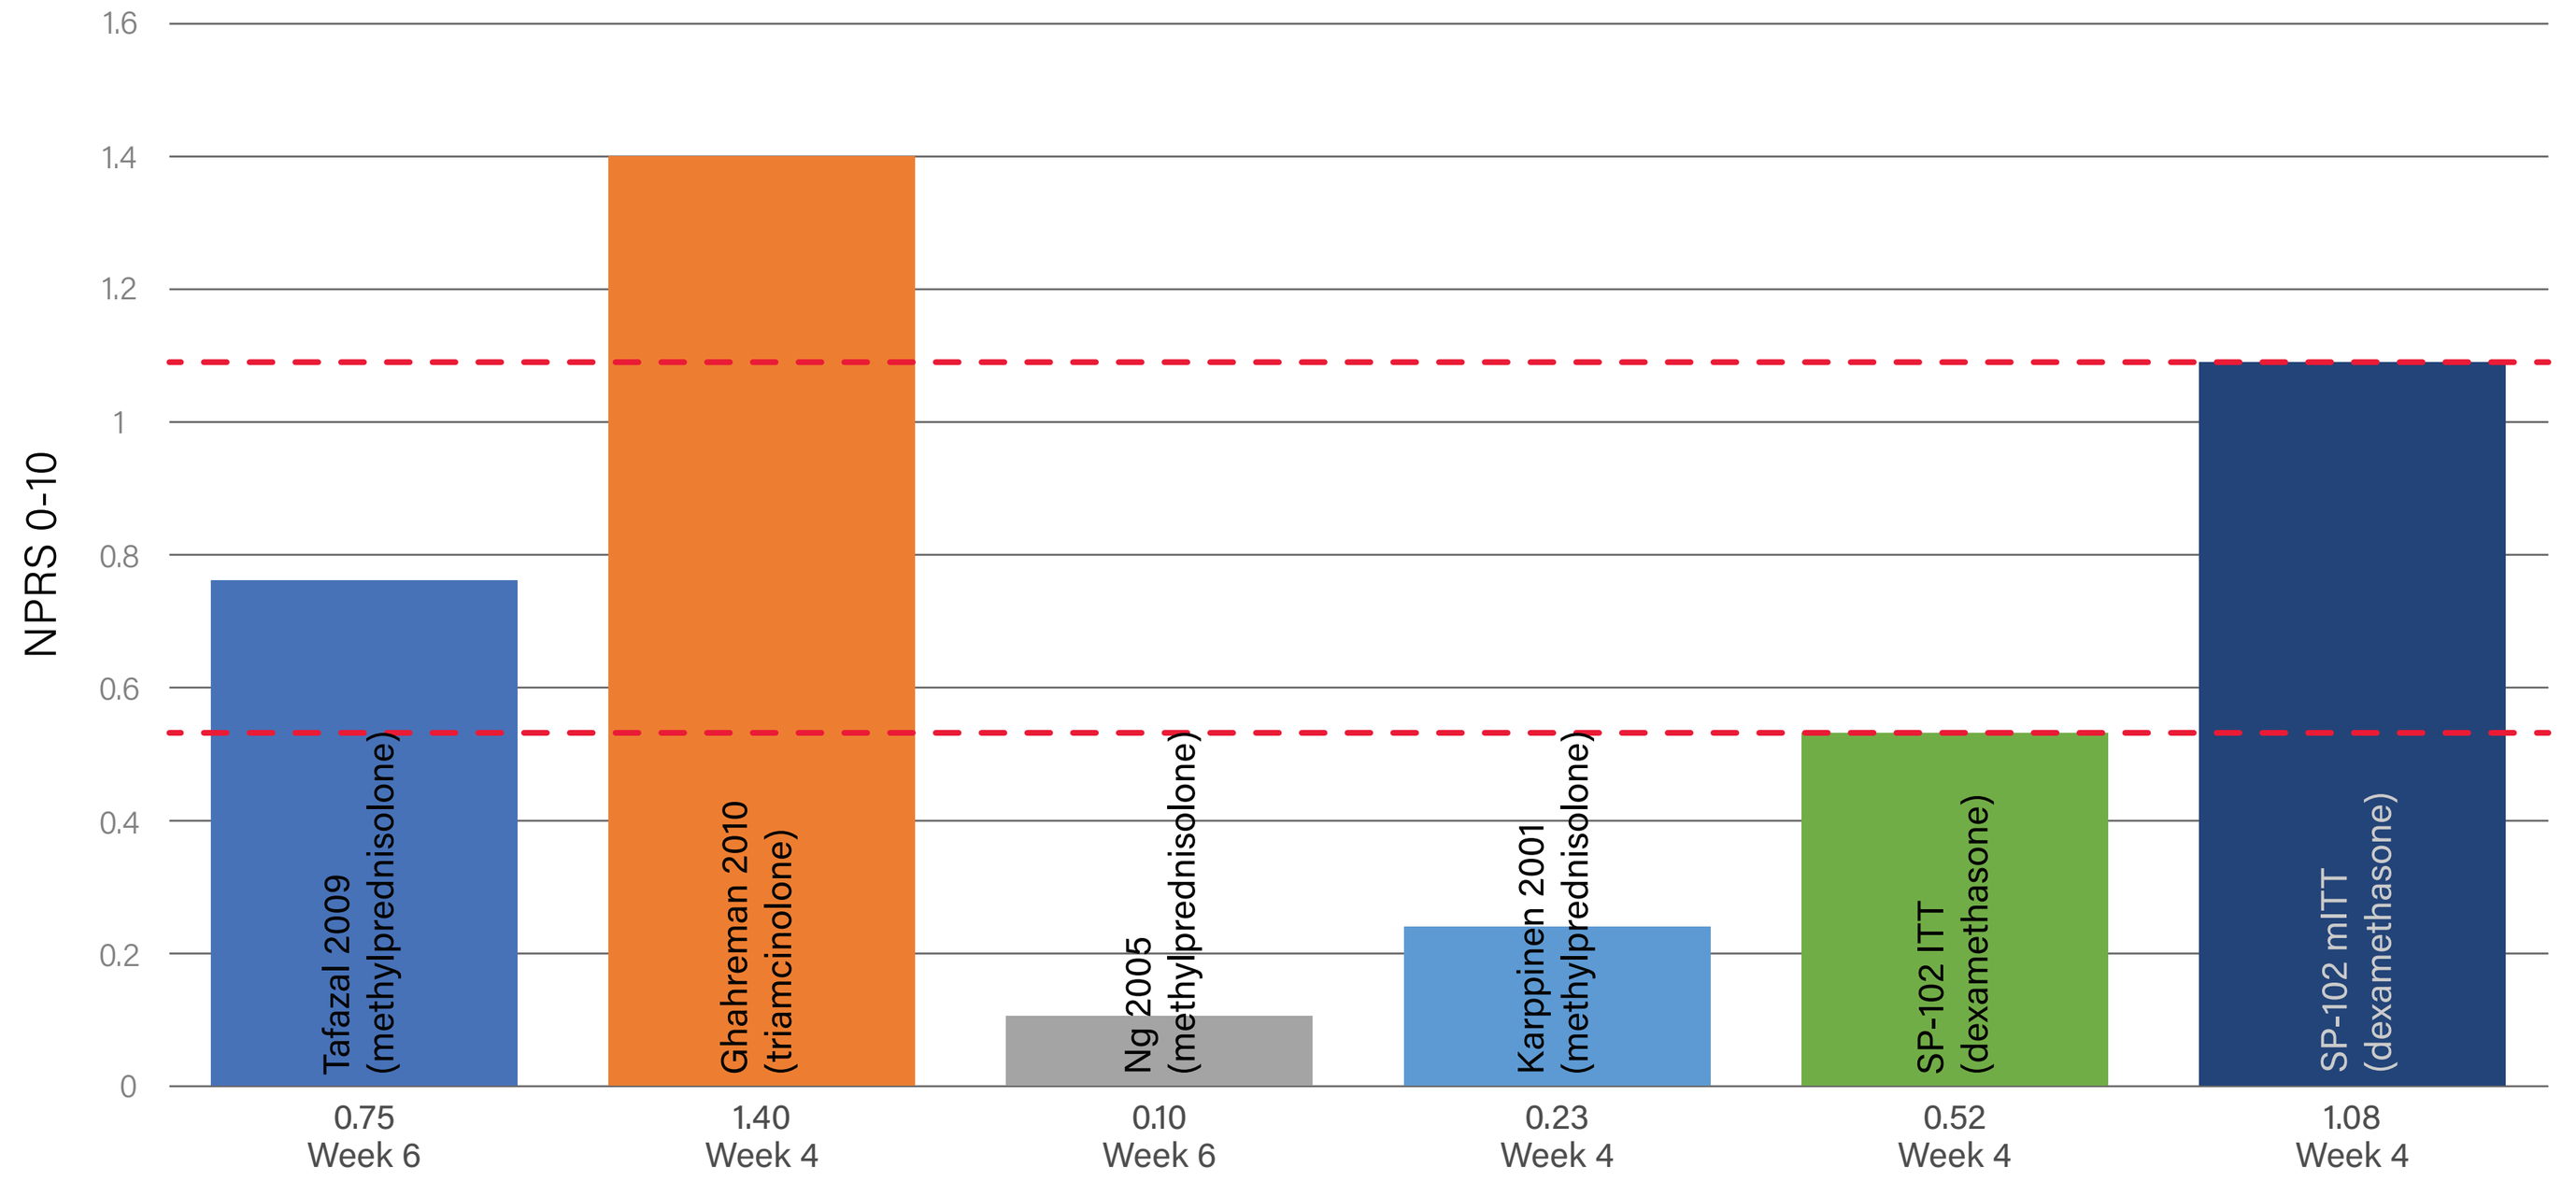
NPRS 0-10
Tafazal 2009 (methylprednisolone)
0.75
Week 6
Ghahreman 2010 (triamcinolone)
1.40
Week 4
Ng 2005 (methylprednisolone)
0.10
Week 6
Karppinen 2001 (methylprednisolone)
0.23
Week 4
SP-102 ITT (dexamethasone)
0.52
Week 4
SP-102 mITT (dexamethasone)
1.08
Week 4

Results for the mITT population (SES = 0.68) for SP-102 produced the highest SES among the studied TFIs (Figure 3A). In the ITT population, the SES (SES = 0.28) was smaller than one study, similar to one study and greater than two studies (Figure 3A). Results were similar when the NPRS GMD were compared with SP-102 showing the second highest separation in the mITT population and 3rd highest for the ITT (Figure 3B).

DISCUSSION

- Interpretation of the clinical meaningfulness of results requires consideration of primary and secondary endpoints assessed with-in subject based on consensus benchmarks, and the between group differences in the context of the overall risk-benefit and in comparison to other approved products for similar indications.
- The results of the CLEAR Trial show clear clinically meaningful separation between SP-102 and placebo in multiple with-in subject endpoints (NPRS Responder Analyses, ODI and BPI Pain Interference). This result is enhanced when solely considering the mITT population.
- This trend is also observed in the magnitude of GMD, particularly when the SESs of SP-102 is compared to other marketed and approved analgesics for CLBP, as well as other TFIs.
- The SP-102 formulation was intentionally created to offer a safer alternative to current off-label use of products that contain warnings of the potential dangerous and life-threatening AEs associated with epidural administration.
- Collectively, the benefit-risk profile presented supports SP-102 being a safe and efficacious product that has the potential to offer a much-needed therapy for the treatment of LRP.

1. Dworkin, R.H., Contemp Clin Trials 2016. 46(11). 2. Smith, S., et al., Pain 2020. 161; S3-S13. 3. Smith, S., et al. Pain 2020; 2446-2461. 4. Dworkin, R., et al. Pain 2009; 238-244. 5. Frymoyer, J.NEJM 1988; 291-300. 6. Frymoyer, J. Instr Course Lect 1992; 217-223. 7. Stafford, M., et al. Br J Anaesth 2007; 461-473. 8. Miller, A., et al., Pain, 2024. 9. Oliveira, C., et al., Cochrane Database of Systematic Reviews 2020. 10. Ostelo, R., et al., Spine 2008; 90-94.