AI-assisted comparison of institutional protocols for regional anesthesia in patients on anticoagulation with ASRA 5th-edition guidance: Multi-institution evaluation

Presenter:
Alex Peres, MD, MSc

Background

Hospitals adapt society guidance into local protocols, but these documents lag and drift, creating variation and cost.

Clinicians lack time to reconcile local text with the latest ASRA recommendations.

We built an AI-enabled audit to flag Aligned / Inconsistent / Missing items at scale.

Methods

96 recommendations × 5 hospitals = 480

Pipeline: extract ASRA + local clauses (LLMs) → retrieval mapping + label (A/I/M) → validate vs anesthesiologist adjudication

Analyses: χ^2 for alignment vs evidence level (IA/IB/IC/IIC/2C) and newness (legacy vs new).

Our **AI audit identifies gaps and inconsistencies** between institutional protocols and ASRA guidelines.

Scan to see the full interactive heatmap

Alignment of Local Protocols With ASRA Recommendations (n=96)

Aligned 40.2% (193) | Inconsistent 22.5% (108) | Missing 37.3% (179)

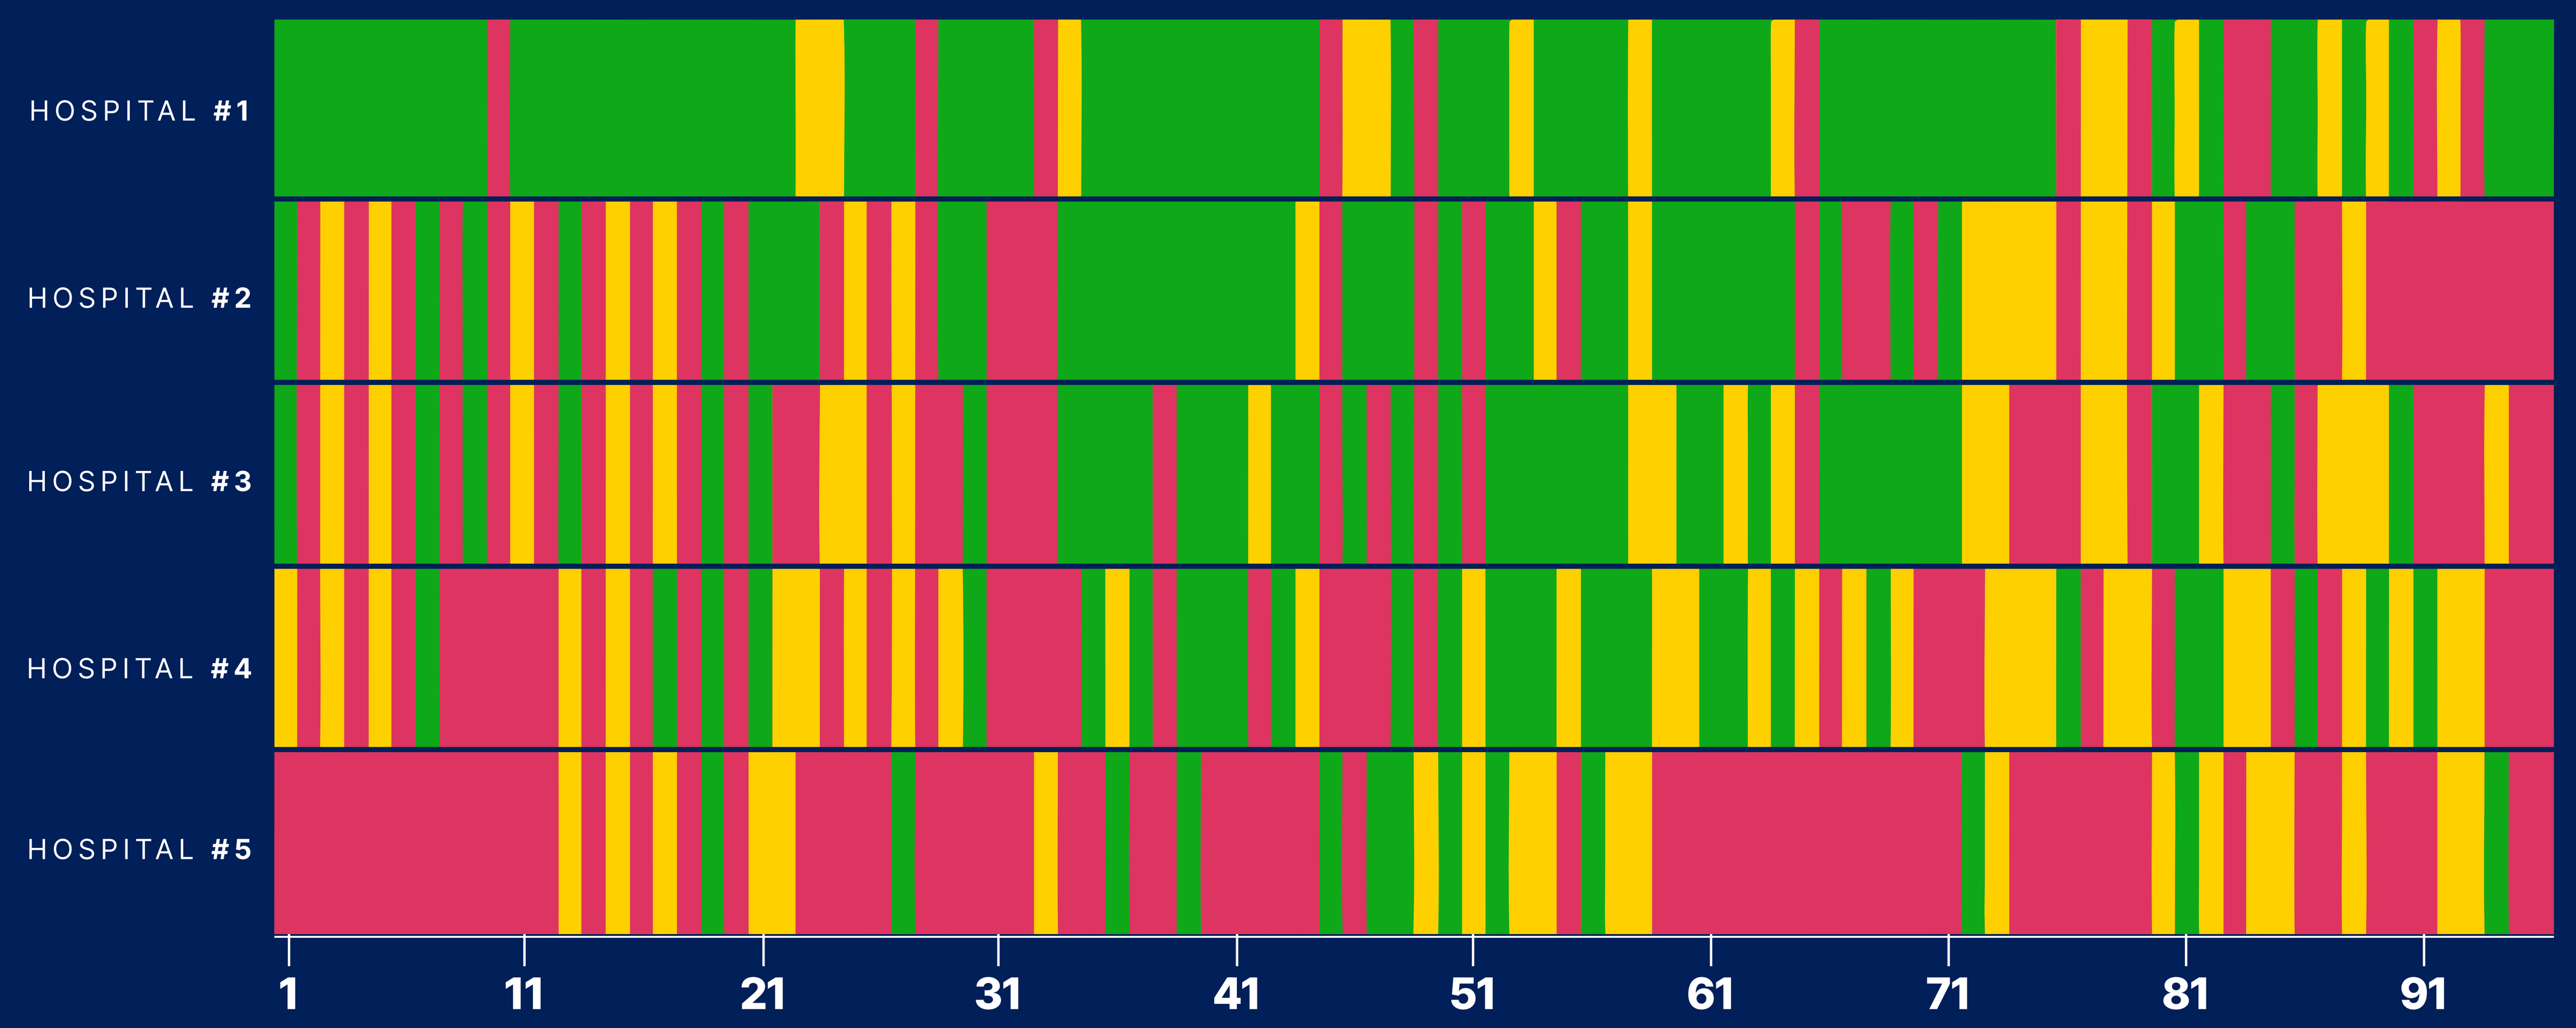

HOSPITAL #1
HOSPITAL #2
HOSPITAL #3
HOSPITAL #4
HOSPITAL #5
1
11
21
31
41
51
61
71
81
91

ASRA RECOMMENDATION INDEX (1-96)

	ALIGNED	INCONS.	MISSING	ALIGN.
HOSPITAL #1	70	14	12	72.9%
HOSPITAL #2	43	21	31	44.8%
HOSPITAL #3	38	22	36	39.6%
HOSPITAL #4	28	31	37	29.2%
HOSPITAL #5	13	20	63	13.5%

C8 Health

ASRA PAIN MEDICINE 51st Annual Regional Anesthesiology and Acute Pain Medicine Meeting
April 16-18, 2026 | Phoenix, Arizona

Key findings & implications

Large cross-site gaps: 59.8% of items were misaligned (37.3% missing; 22.5% inconsistent).

Reliable audit vs anesthesiologists' adjudication: ~93% accuracy, ~93% precision, ~98% recall.

Where gaps concentrate: Lower alignment for new and lower level of evidence items (new 20% vs legacy 51.5% aligned; IA 72.5% aligned vs. IC 53.8% and IIC 34.2%, χ^2 $p<0.001$).

The model identifies not only where institutions lag behind current guidelines, but also local practices not yet formally adopted by society, flagging these as candidates for future guidelines updates.

Alex Peres, MD, MSc[1], Sahil Amin, MD[2], Tomer Cramer, PhD[3], Ido Zamberg, MD, MEHP[3]

Department of Anesthesiology, Montefiore Medical Center, Albert Einstein College of Medicine, The Bronx, NY, USA[1]

Department of Anesthesiology, Critical Care and Pain Medicine, Beth Israel Deaconess Medical Center, Boston, MA, USA[2]

C8 Health Inc, New York, New York, USA[3]