Evaluating the use of sublingual sufentanil in patients with buprenorphine treatment who are undergoing ambulatory surgery: A Prospective Case Series

Authors: Kateryna Slinchenkova[1] BA, Elilary Montilla Medrano[2] MD, Adam S. Yedlin[2] MD, Preeti Anand[2] MD, Karina Gritsenko[2] MD, Sofia Steinberg[2] DO, Kathryn Breidenbach[2] MD, Sarang Koushik[3] MD, Naum Shaparin[2] MD

Affiliations: 1. New York Medical College School of Medicine, Valhalla, NY, USA 2. Department of Anesthesiology, Montefiore Medical Center, Albert Einstein College of Medicine, Bronx, NY, USA 3. Department of Anesthesiology, Valleywise Health Medical Center, Creighton University School of Medicine, Phoenix, AZ, USA

49th Annual Regional Anesthesiology and Acute Pain Medicine Meeting
March 21-23, 2024 | San Diego, CA
ASRA PAIN MEDICINE

Background

One of the long-term management options to avoid opioid use relapse is to include an opioid agonist (methadone or buprenorphine) in the management regime. (1,2) The number of Medicaid-covered prescriptions for buprenorphine containing products for treatment has increased 5-fold from 2011 to 2018. (3) Since the guideline change in 2021, providers no longer need additional training or licensure to prescribe buprenorphine, so prescription numbers and access to this medication have an expectation to increase further. Recommendations for perioperative treatment of patients on suboxone have varied in recent years, with many discussions on a need for discontinuation or tapering of dose due to the potential for pain control difficulties. (4) Recent guidelines have pivoted to suggest that buprenorphine should be continued and postoperative pain management can be achieved with the focused utilization of regional anesthesia and IV non-opioid anesthesia, with opioids acting as a supplement. Nevertheless, conversion to methadone or morphine is noted as an option to avoid difficulties.(5) A multimodal approach to analgesia and communication with prescribing provider is recommended. (6) There is a known concern that increased IV medication and increased opioid need to achieve pain control can lead to adverse effects such as nausea, respiratory depression, and more. Furthermore, the need for this intervention may increase patient length of stay and require increased monitoring. One potential medication that may assist with such issues is sufentanil.

Overall, a literature review of the sublingual sufentanil tablet (SST) found it to be an excellent treatment for chronic pain patients, reducing the need for IV medication. (7) In non-opioid tolerant patients, it has shown great success. Two randomized trials of different SST dosages showed that it significantly reduced pain intensity. Both studies indicated an increased rate of nausea for those not in the placebo group. (8, 9) Lower PACU opioid administration was noted (10) and those receiving sublingual sufentanil experienced less oxygen desaturation below 95% (p=0.028). (11) No research is currently available on the efficacy of sublingual sufentanil in opioid-tolerant patients.

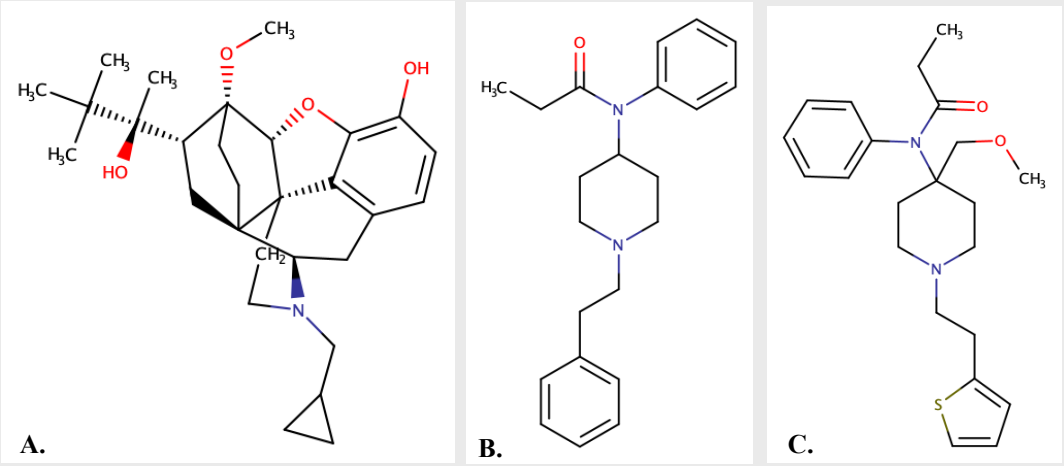
A.
B.
C.

Figure 1: *Structures of buprenorphine (A), fentanyl (B), and sufentanil (C).* Out of the commonly used opioids, sufentanil is the only one that has a higher affinity (K_i =0.138) the mu receptor than buprenorphine (K_i =0.2157). It also outcompetes hydromorphone, morphine, and fentanyl (K_i =0.3654,1.168, 1.346, respectively). (12) Images created in Marvin Sketch 23.14.

Methods And Materials

Patients were screened using a weekly patient report of upcoming surgeries. Patients meeting the following criteria were approached for study participation:

Inclusion Criteria	Exclusion Criteria
Aged 18-100	Known allergic reactions to sublingual sufentanil
Currently taking buprenorphine or buprenorphine containing medication for opioid use disorder	Severe respiratory illness
ASA Physical Score 1-3	Significant intraoperative hemodynamic instability
Surgery done under general anesthesia	Use of regional Anesthesia techniques
Able to provide signed informed consent	

Table 1. Inclusion and Exclusion criteria for study enrollment

Patients meeting criteria were called prior to surgery to offer participation in the study. Informed consent discussion was done over phone and written consent was obtained the morning of surgery.

The study was IRB-approved by Einstein IRB and sponsored by AcelRx Pharmaceuticals as an investigator-initiated trial. All three patients enrolled received 975mg acetaminophen preoperatively, and standardized general anesthesia, as per site protocol. The sublingual sufentanil tablet was administered by the attending anesthesiologist immediately following induction of general anesthesia.

Patients were evaluated for adverse effects upon entering recovery, an hour after recovery start, and 24 hours after discharge.

Cases

	Patient 1	Patient 2	Patient 3
Age, Sex	58, M	52, F	45, F
Daily Suboxone Dosage (mg)	4	8	12
Procedure Name	Prostatic urethral lift	Bilateral breast reduction mammoplasty	Vaginal sling for stress incontinence
Procedure Length	10 minutes	4 hours, 36 minutes	59 minutes
PACU Length of Stay (hours)	1.5	2.5	2.25
Intraoperative Analgesics Administered	none	100 mcg fentanyl 75 mcg of 2% lidocaine	100 mcg fentanyl 100mg of 2% lidocaine
Post-op Analgesics Administered	none	none	30 mcg sublingual sufentanil* 800 mg IV ibuprofen*
Medications Prescribed for Discharge	Acetaminophen, PRN	Acetaminophen, PRN	Acetaminophen PRN 5 mg immediate release oxycodone tablet, 6 times a day
Pain score upon PACU arrival	0/10	0/10	8/10
Pain score an hour after PACU arrival	0/10	0/10	2/10*
Pain Score 24 hours after discharge	0/10	No contact made	5/10**

Table 2: *Summary of case facts.* In addition to the above-mentioned medication, all patients received 30 mcg intraoperatively. All patients had no missed doses of buprenorphine. No adverse events were reported.

*Patient 3 reported an initial pain of 8/10 but within half an hour, it increased to a 10/10. Starred medication were administered. Within half an hour, the pain subsided to a 2/10.
**Patient reported that pain medicine was not ineffective when completing follow up call despite taking both acetaminophen and oxycodone as prescribed.

Discussion

While the results of the 3 cases cannot be taken as conclusive evidence of sublingual sufentanil efficacy in opioid-tolerant patients, it demonstrates an important option for pain management for patients on varying doses of suboxone. All three patients treated had no adverse effects and tolerated recovery room pain well. It is worth noting that in the patient who experienced the most pain, sufentanil administration was effective in lowering the pain from 10/10 to a 2/10. When completing a follow up call, after sufentanil has been cleared, pain control was no longer adequate. The contrasting minimal need for analgesia is novel due to numerous previous case reports reporting a high level of difficulty and complication when treating patients on buprenorphine.

Case Report	**Notable Findings regarding difficult post-operative pain control**
McCormick, et. al. (13)	Required hydromorphone PCA at 0.8 mg with a 15 minute lockout. Patient was transferred to oral opioids after pain plateaued at 3/10 2 days after surgery. Suboxone use had to be stopped.
*Huang, et. al. (*14)	Over 70 mg IV hydromorphone as well as oral opioids were needed to keep pain "tolerable". Complete pain control was not successful until buprenorphine was stopped.
Brummett, et. al. (15)	High dose of opioids and dexmedetomidine therapy needed after spine revision surgery. At-rest pain was controlled but patient experienced 5/10 pain out of bed and had to be closely monitored in the ICU.
Chern, et. al. (16)	When stopping buprenorphine and transitioning to hydromorphone 5 days before removal of vaginal mesh and cystoscopy, a multimodal approach was still needed for controlled pain.

Table 3. Notable case reports on pain management of patients on buprenorphine treatment.

While it is difficult to compare the expected pain from the varied procedures discussed in reports, the end results are worth noting. Pain management using sublingual sufentanil is fast, effective, and minimally invasive allowing for repeat dosing if needed. Its pharmacological profile allows for doses to be kept low, which limits the risk of adverse events such as respiratory depression. The treatment of acute pain plays a major role in preventing chronic pain development (17) While the patients on buprenorphine are already being treated for pain, preventing further chronic pain development in addition to their baseline helps keep medication doses constant and prevent quality of life changes.

Overall, further research is needed to attest for the efficacy of sublingual sufentanil in opioid-tolerant patients. However, the case report presented speaks for the benefit of individual consideration and implementation due to excellent pain management and lack of adverse events seen.

Conclusions

- Due to buprenorphine's high affinity for the mu receptor, common opioids such as morphine or fentanyl are needed in higher doses in the case of postoperative pain management. Sufentanil has a higher affinity and can be administered in lower doses.
- In three patients undergoing surgery, sublingual sufentanil was effective in controlling pain. When sufentanil effects wore off in one patient, pain management was no longer effective.
- Although further research is needed to verify validity, sufentanil may be a viable alternative in patients on buprenorphine for postoperative pain management

References

1. R.E. Johnson, P.J. Fudala, R. Payne, Buprenorphine: considerations for pain management, Journal of pain and symptom management. 29 (2005) 3 297-326.
2. D.P. Alford, P. Compton, J.H. Samet. Acute pain management for patients receiving maintenance methadone or buprenorphine therapy. American College of Physicians; 2006.
3. https://www.urban.org/sites/default/files/publication/100817/2019.08.19_av_state_medicaid_rx_oud_final_v3_4.pdf
4. T. Anthony Anderson, Aurora N. A. Quaye, E. Nalan Ward, Timothy E. Wilens, Paul E. Hilliard, Chad M. Brummett; To Stop or Not, That Is the Question: Acute Pain Management for the Patient on Chronic Buprenorphine. *Anesthesiology* 2017; 126:1180–1186 doi: https://doi.org/10.1097/ALN.0000000000001633
5. Roberts, D. M., & Meyer-Witting, M. (2005). High-dose buprenorphine: perioperative precautions and management strategies. *Anaesthesia and intensive care*, *33*(1), 17–25. https://doi.org/10.1177/0310057X0503300104
6. Champagne, K., Date, P., Forero, J. P., Arany, J., & Gritsenko, K. (2022). Patients on Buprenorphine Formulations Undergoing Surgery. *Current pain and headache reports*, *26*(6), 459–468. https://doi.org/10.1007/s11916-022-01046-6
7. Koushik, S. S., Schwartz, R. H., Cherkalin, D., Sankar, V., Shaparin, N., & Viswanath, O. (2022). A Review of Sublingual Sufentanil Tablet (SST) and its Utility as an Analgesic Agent for Pain Procedures. *Current pain and headache reports*, *26*(2), 145–149. https://doi.org/10.1007/s11916-022-01014-0
8. Minkowitz, H. S., Leiman, D., Melson, T., Singla, N., DiDonato, K. P., & Palmer, P. P. (2017). Sufentanil Sublingual Tablet 30 mcg for the Management of Pain Following Abdominal Surgery: A Randomized, Placebo-Controlled, Phase-3 Study. *Pain practice : the official journal of World Institute of Pain*, *17*(7), 848–858. https://doi.org/10.1111/papr.12531
9. Jove, M., Griffin, D. W., Minkowitz, H. S., Ben-David, B., Evashenk, M. A., & Palmer, P. P. (2015). Sufentanil Sublingual Tablet System for the Management of Postoperative Pain after Knee or Hip Arthroplasty: A Randomized, Placebo-controlled Study. *Anesthesiology*, *123*(2), 434–443. https://doi.org/10.1097/ALN.0000000000000746
10. Seify H. (2022). Sufentanil Sublingual Tablet Reduces Postoperative Opioid Use Following Outpatient Plastic Surgery. *Aesthetic surgery journal. Open forum*, *4*, ojac040. https://doi.org/10.1093/asjof/ojac040
11. Melson, T.I., Boyer, D.L., Minkowitz, H.S., Turan, A., Chiang, Y.-K., Evashenk, M.A. and Palmer, P.P. (2014), Sufentanil Sublingual Tablet System vs. Intravenous Patient-Controlled Analgesia with Morphine for Postoperative Pain Control: A Randomized, Active-Comparator Trial. Pain Pract, 14: 679-688. https://doi.org/10.1111/papr.12238
12. Volpe, D. A., McMahon Tobin, G. A., Mellon, R. D., Katki, A. G., Parker, R. J., Colatsky, T., Kropp, T. J., & Verbois, S. L. (2011). Uniform assessment and ranking of opioid μ receptor binding constants for selected opioid drugs. *Regulatory toxicology and pharmacology : RTP*, *59*(3), 385–390. https://doi.org/10.1016/j.yrtph.2010.12.007
13. McCormick, Z., Chu, S. K., Chang-Chien, G. C., & Joseph, P. (2013). Acute pain control challenges with buprenorphine/naloxone therapy in a patient with compartment syndrome secondary to McArdle's disease: a case report and review. *Pain medicine (Malden, Mass.)*, *14*(8), 1187–1191. https://doi.org/10.1111/pme.12135
14. Huang, A., Katznelson, R., de Perrot, M., & Clarke, H. (2014). Perioperative management of a patient undergoing Clagett window closure stabilized on Suboxone® for chronic pain: a case report. *Canadian journal of anaesthesia = Journal canadien d'anesthesie*, *61*(9), 826–831. https://doi.org/10.1007/s12630-014-0193-y
15. Brummett, C. M., Trivedi, K. A., Dubovoy, A. V., & Berland, D. W. (2009). Dexmedetomidine as a novel therapeutic for postoperative pain in a patient treated with buprenorphine. *Journal of opioid management*, *5*(3), 175–179. https://doi.org/10.5055/jom.2009.0018
16. Chern, S. Y., Isserman, R., Chen, L., Ashburn, M., & Liu, R. (2013). Perioperative Pain Management for Patients on Chronic Buprenorphine: A Case Report. *Journal of anesthesia & clinical research*, *3*(250), 1000250. https://doi.org/10.4172/2155-6148.1000250
17. McGreevy, K., Bottros, M. M., & Raja, S. N. (2011). Preventing Chronic Pain following Acute Pain: Risk Factors, Preventive Strategies, and their Efficacy. *European journal of pain supplements*, *5*(2), 365–372. https://doi.org/10.1016/j.eujps.2011.08.013

Poster Design & Printing by Graphic Arts Center