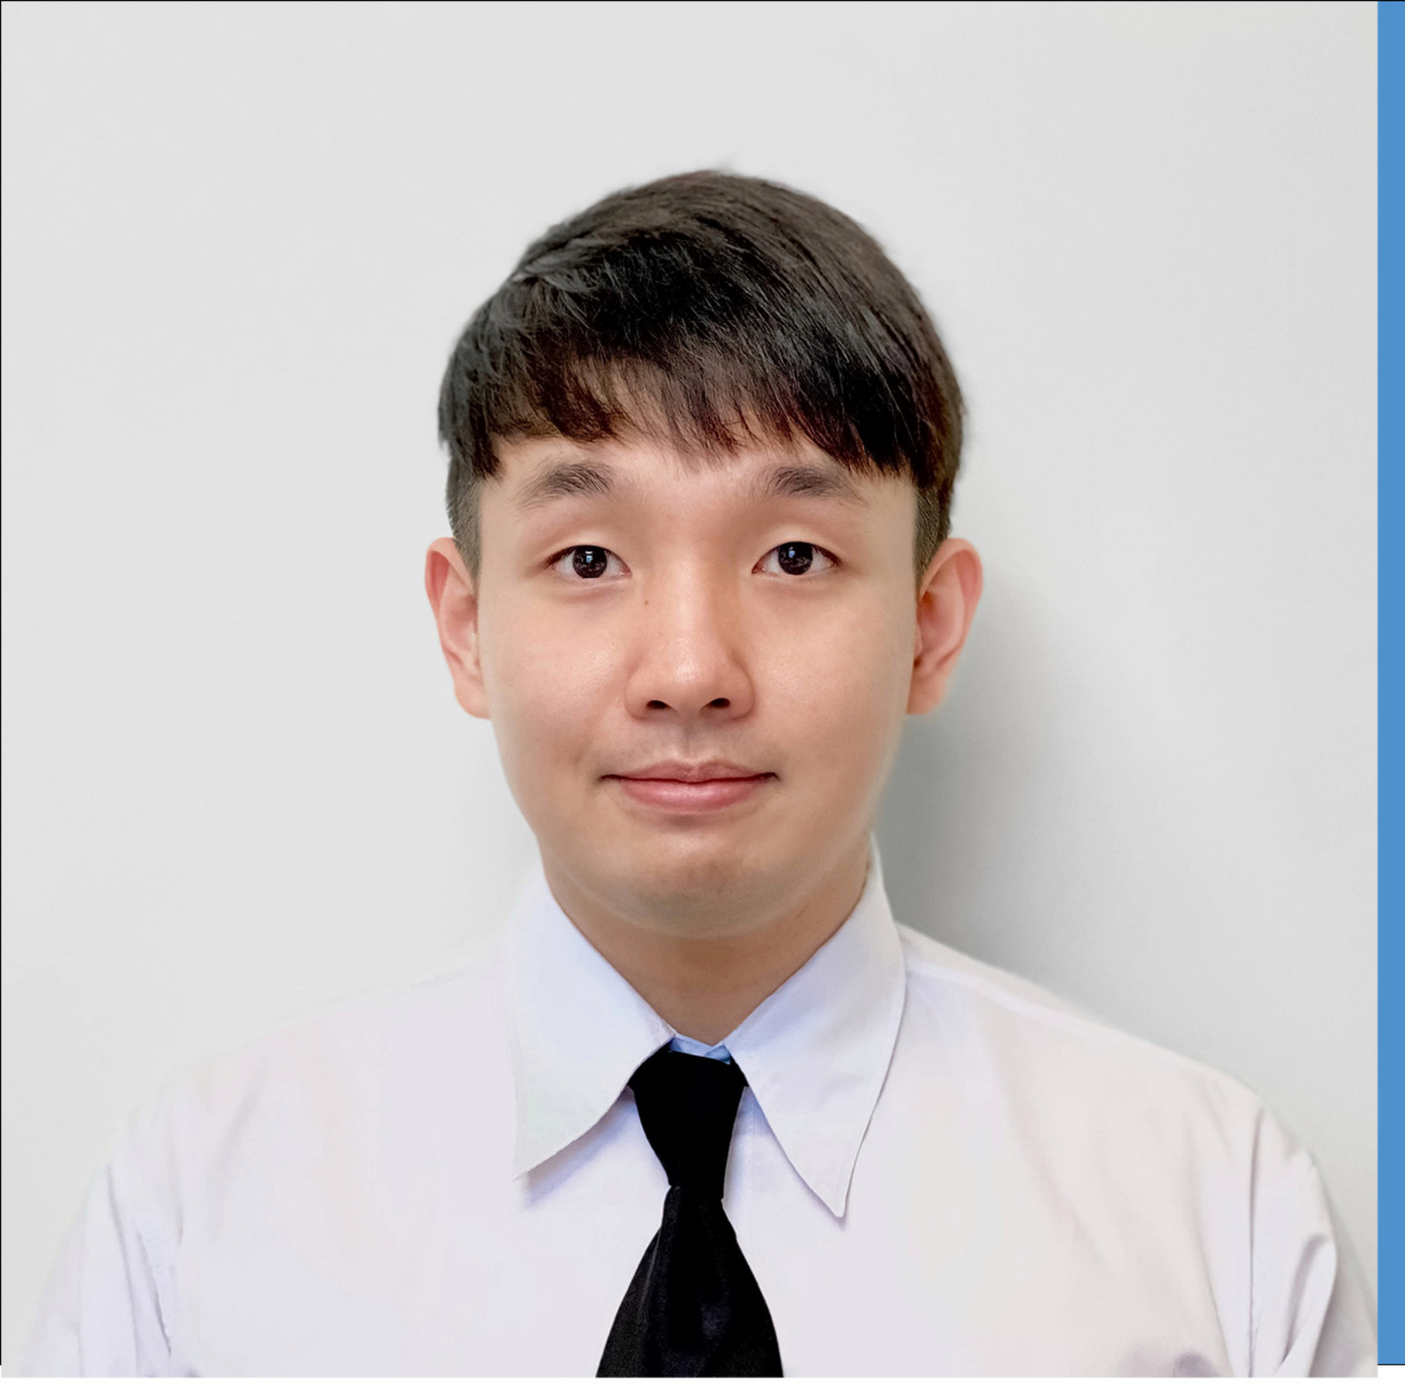

Utilization of a Novel Digital Smile Design Software for Full-Mouth Rehabilitation of a Severely Worn Dentition: A Clinical Report

Jirat Yongsawasdigul DDS, Mohammed Alshaye DMD, Vincent Basha DMD, FACP,
Khaled El Rafie DMD, FACP, Konstantinos Vazouras DMD, DDS, MPhil, MDSc, FACP

Tufts School of Dental Medicine

Objective

This case report demonstrates a modern, esthetically driven approach to full-mouth rehabilitation in a young patient with severe tooth wear, utilizing digital platform for digital smile design and a guided crown lengthening guide to optimize gingival architecture prior to prosthetic treatment.

Methods

A 33-year-old male presented with severe tooth wear, affecting esthetics and occlusal function. Comprehensive digital records were imported into the digital platform to create an ideal smile design. The digital plan facilitated interdisciplinary collaboration and received patient approval. A surgical guide for esthetic crown lengthening was fabricated from the digital wax-up, allowing precise gingival recontouring aligned with the planned restorations. After healing, full-coverage restorations were delivered to reestablish vertical dimension, esthetics, and occlusal stability.

Results & Conclusion

This case highlights the value of integrating digital smile and guided crown lengthening in the full-mouth rehabilitation of a young patient with severe tooth wear. Digital planning enhanced accuracy, efficiency, and interdisciplinary coordination, resulting in a predictable and esthetically pleasing outcome.

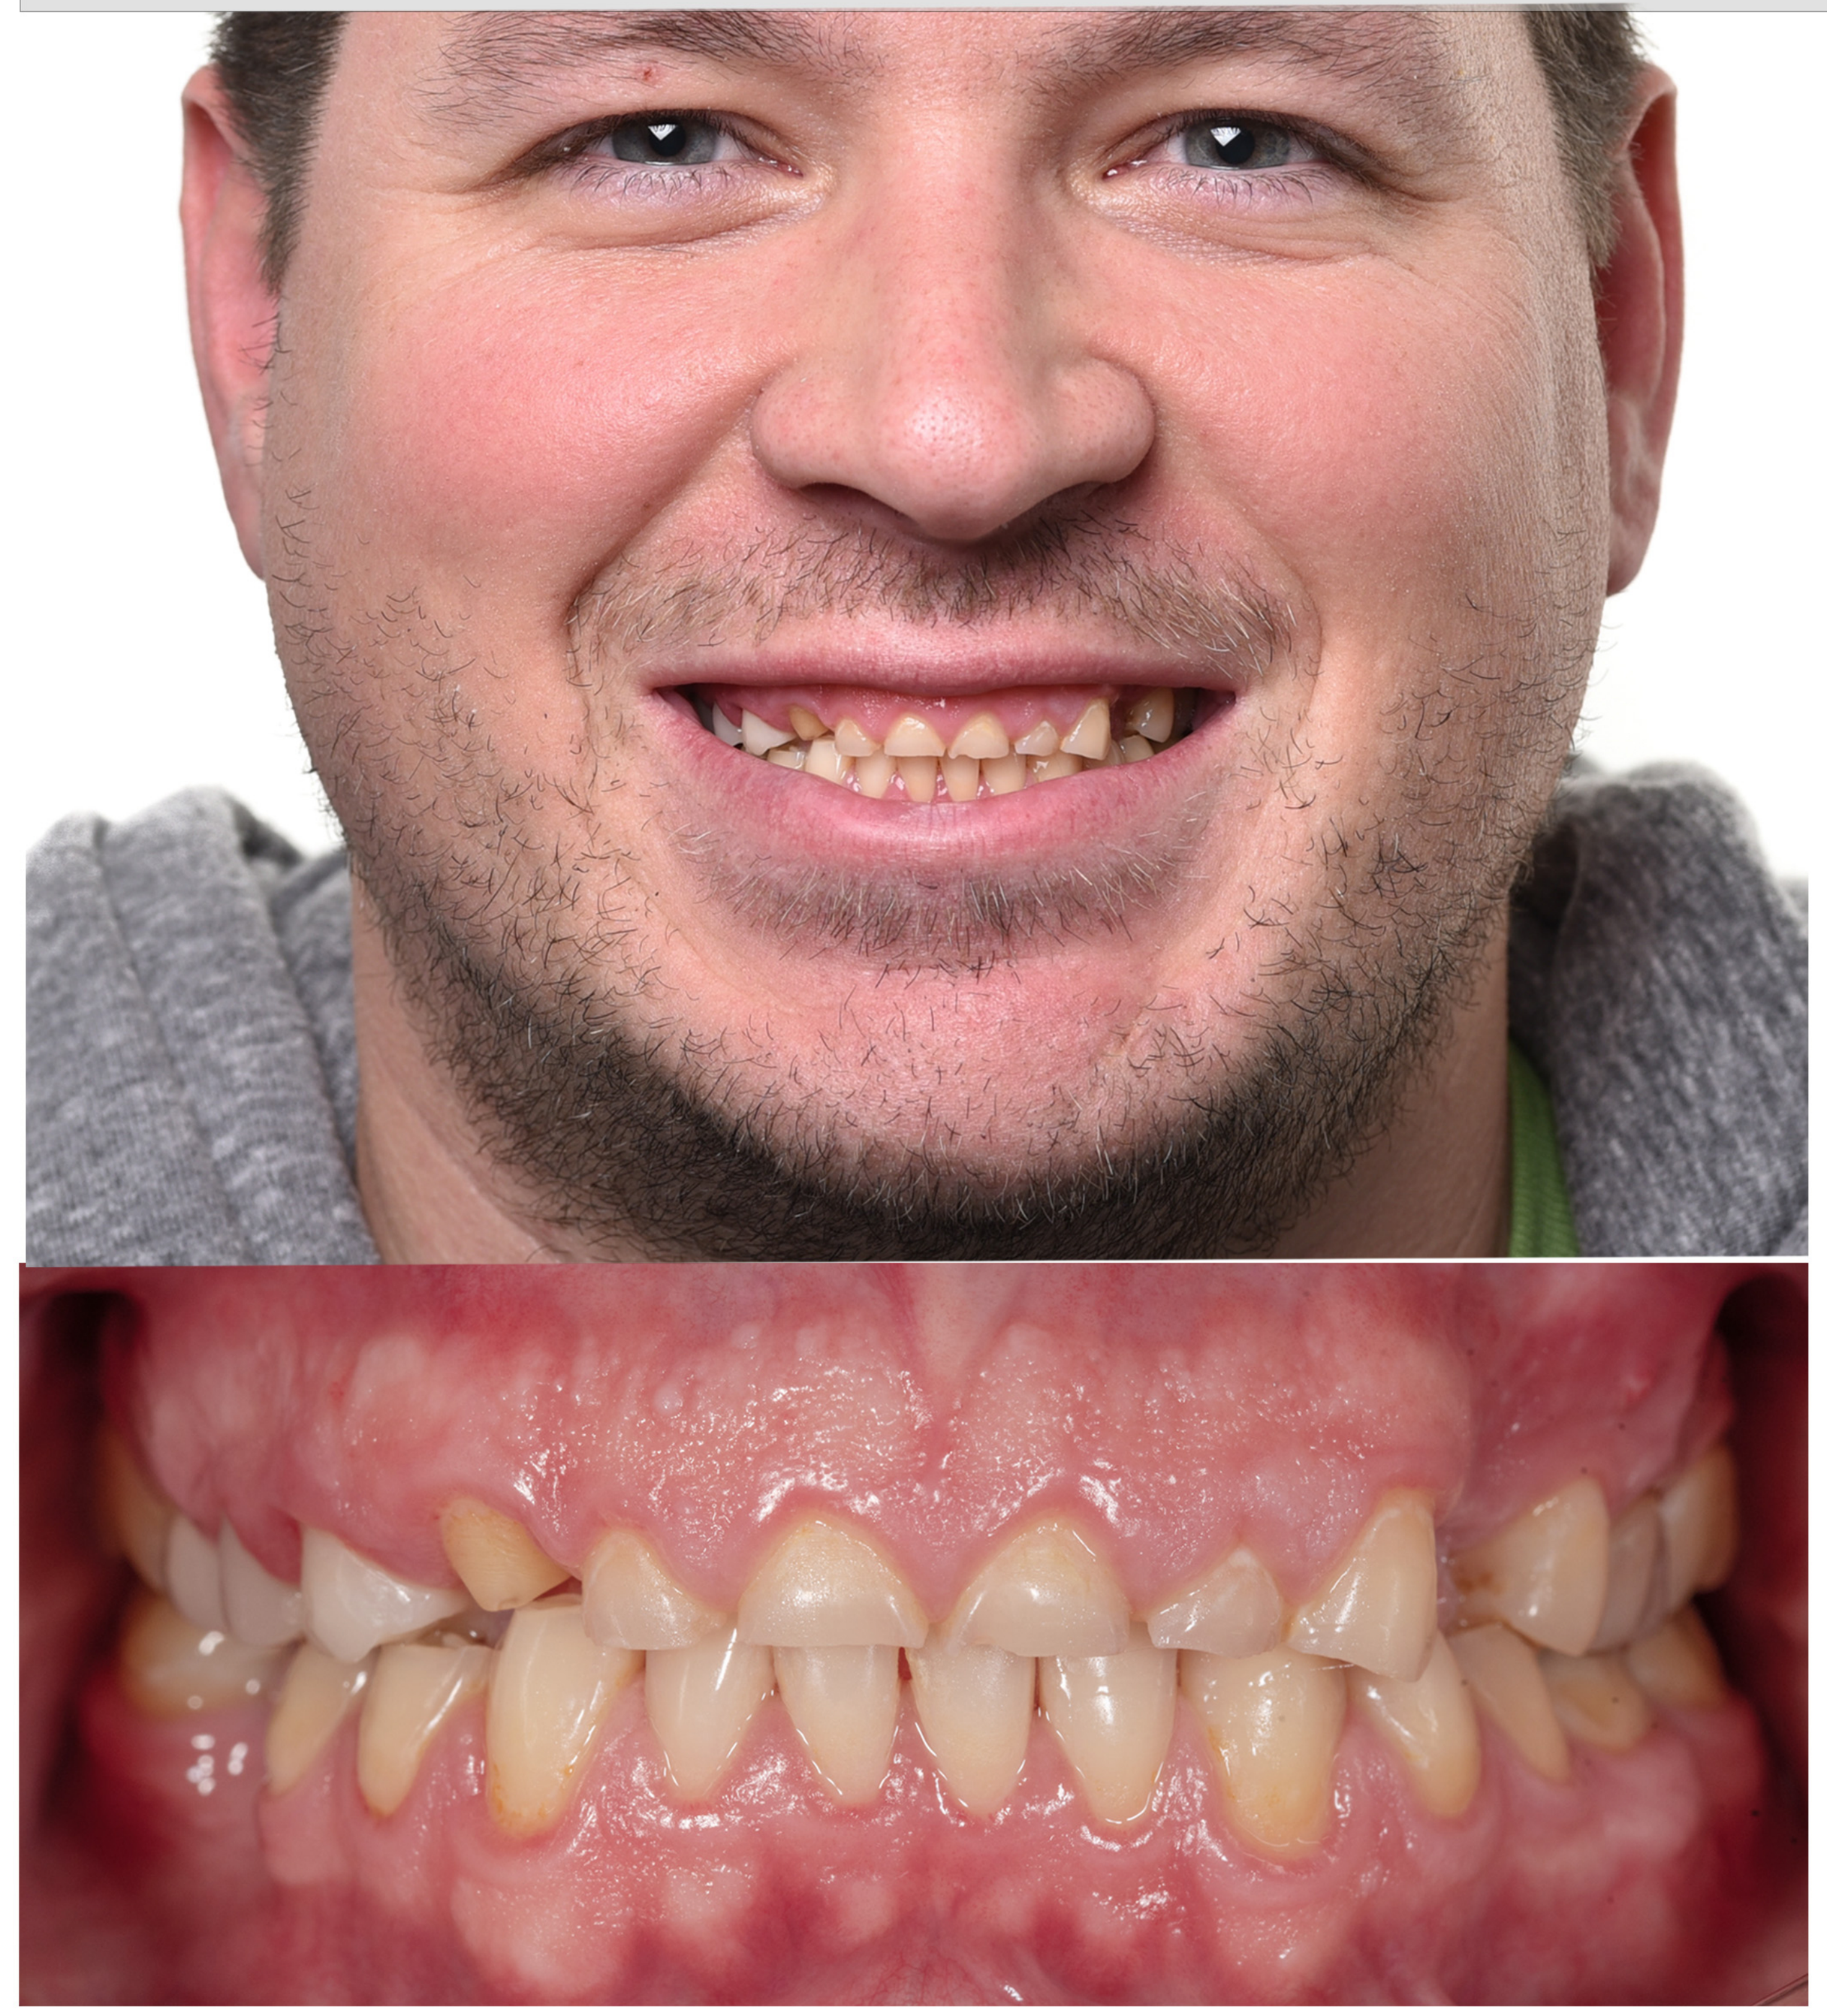

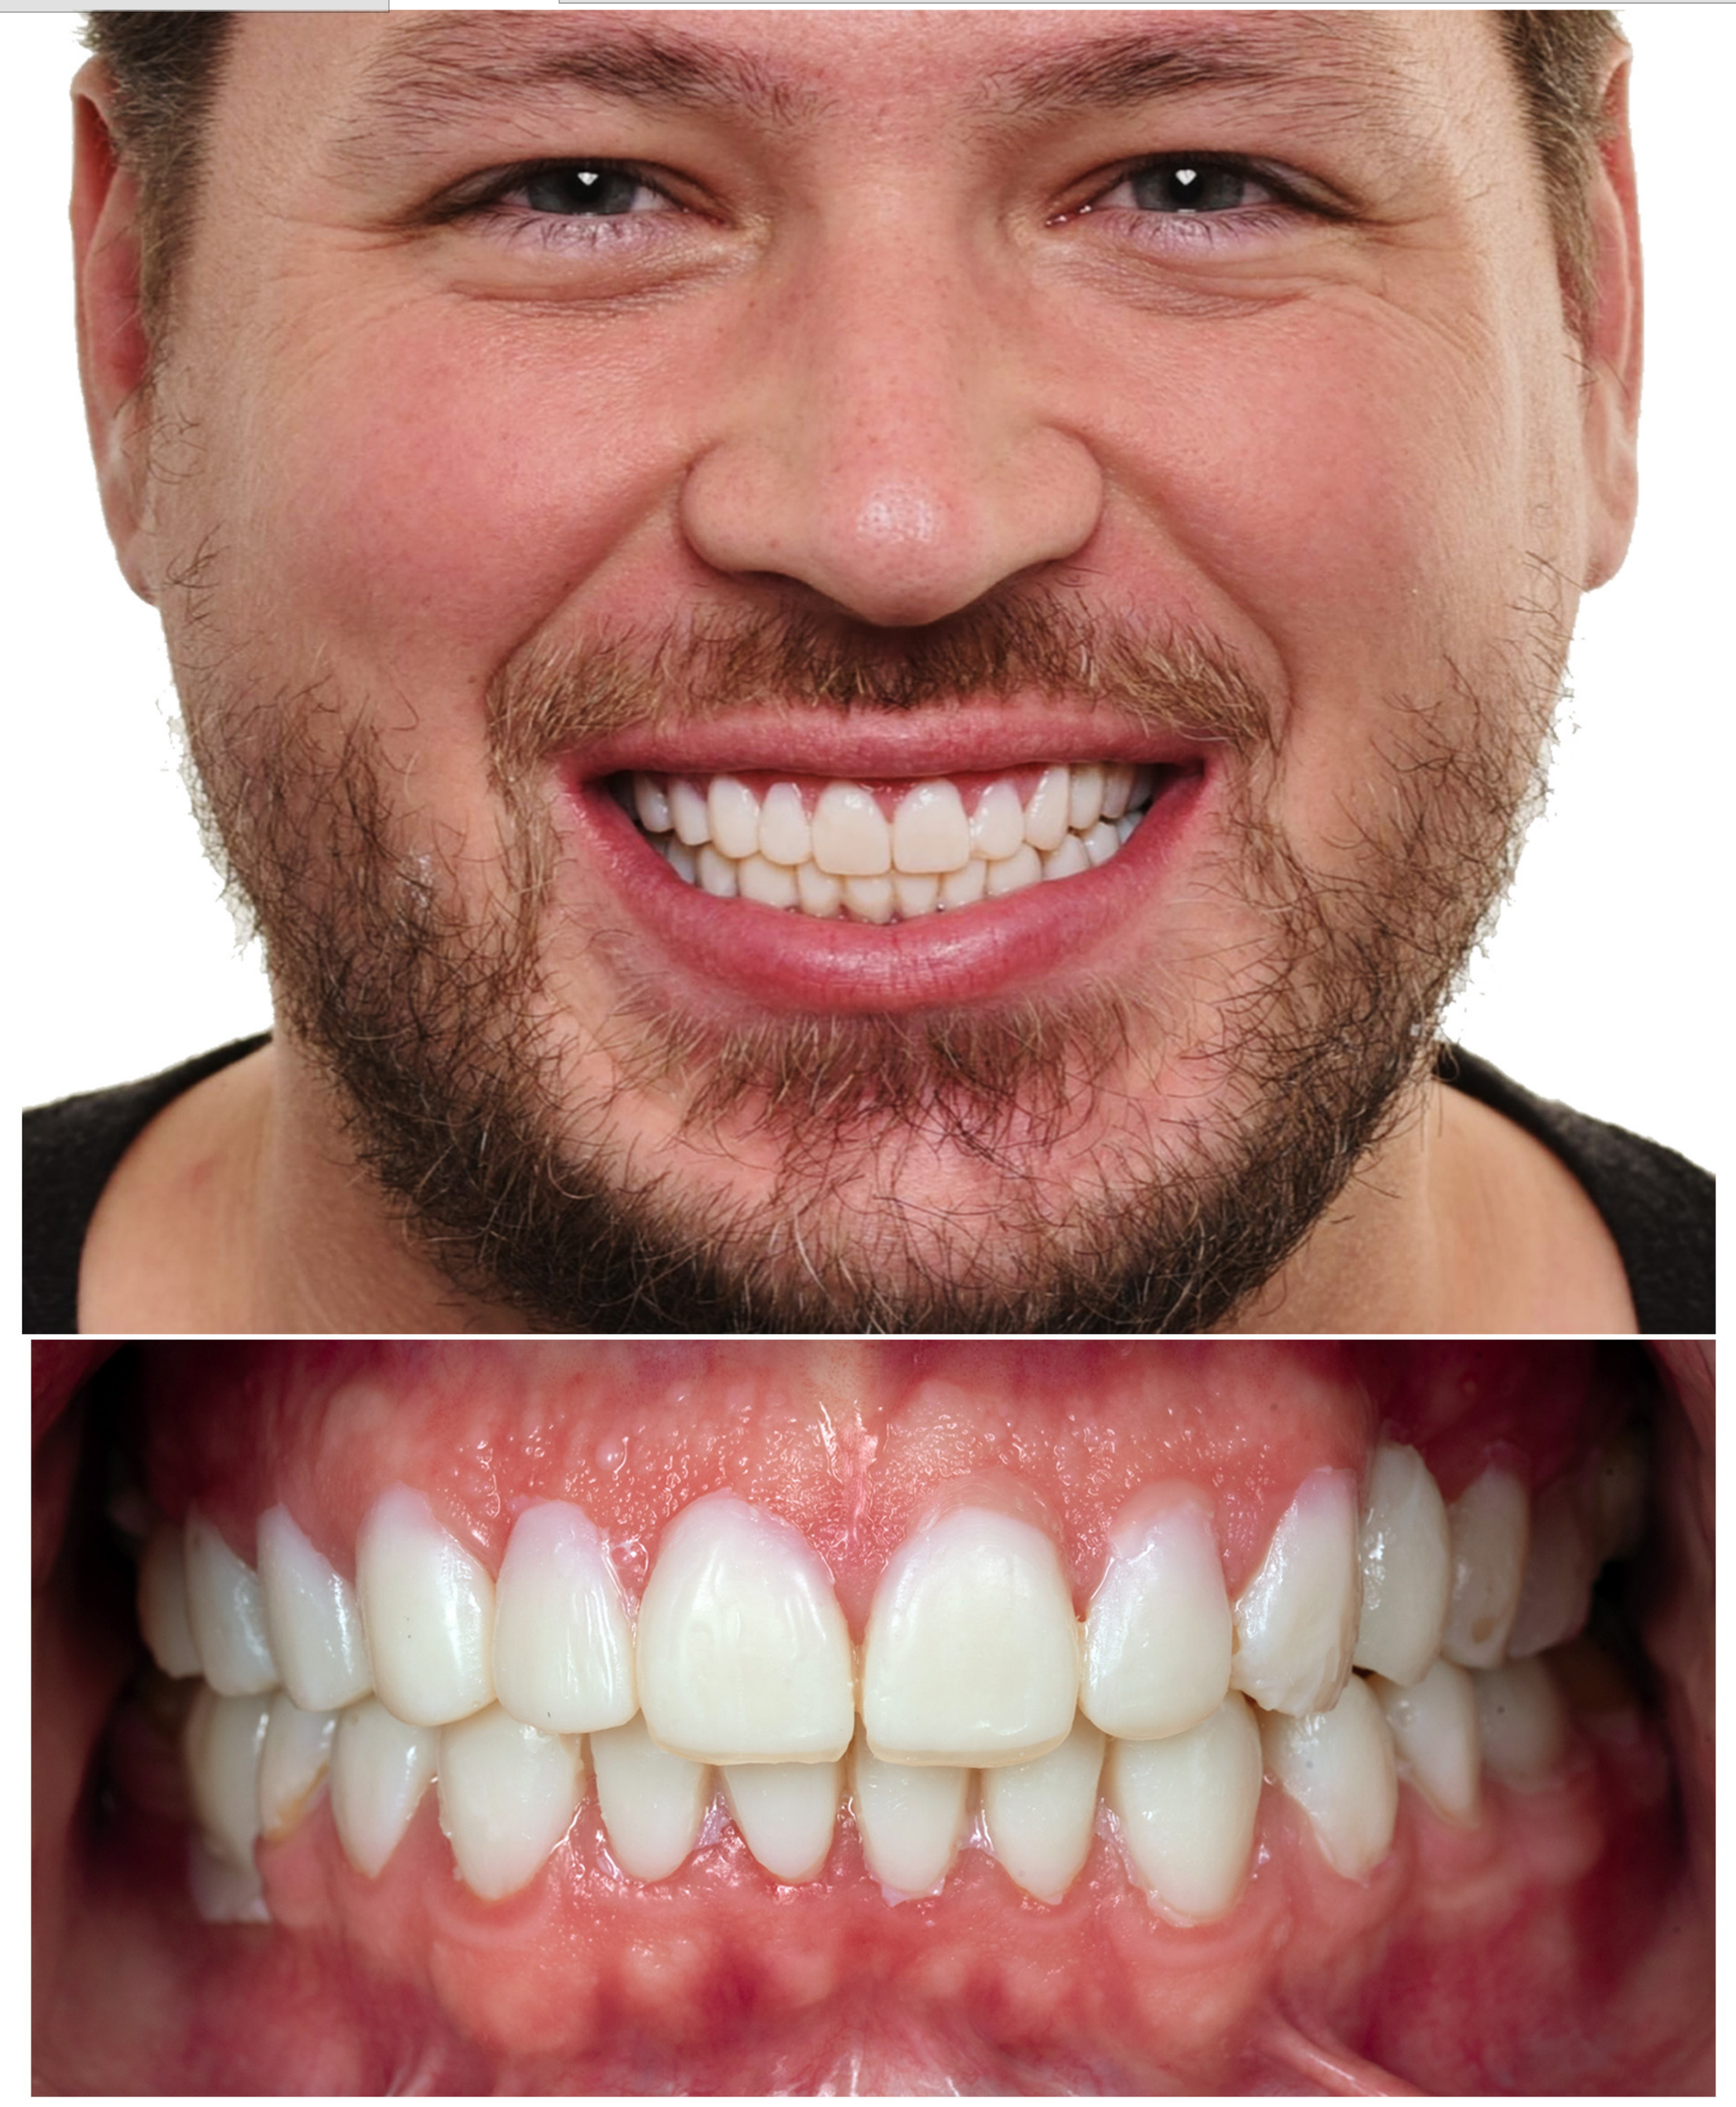

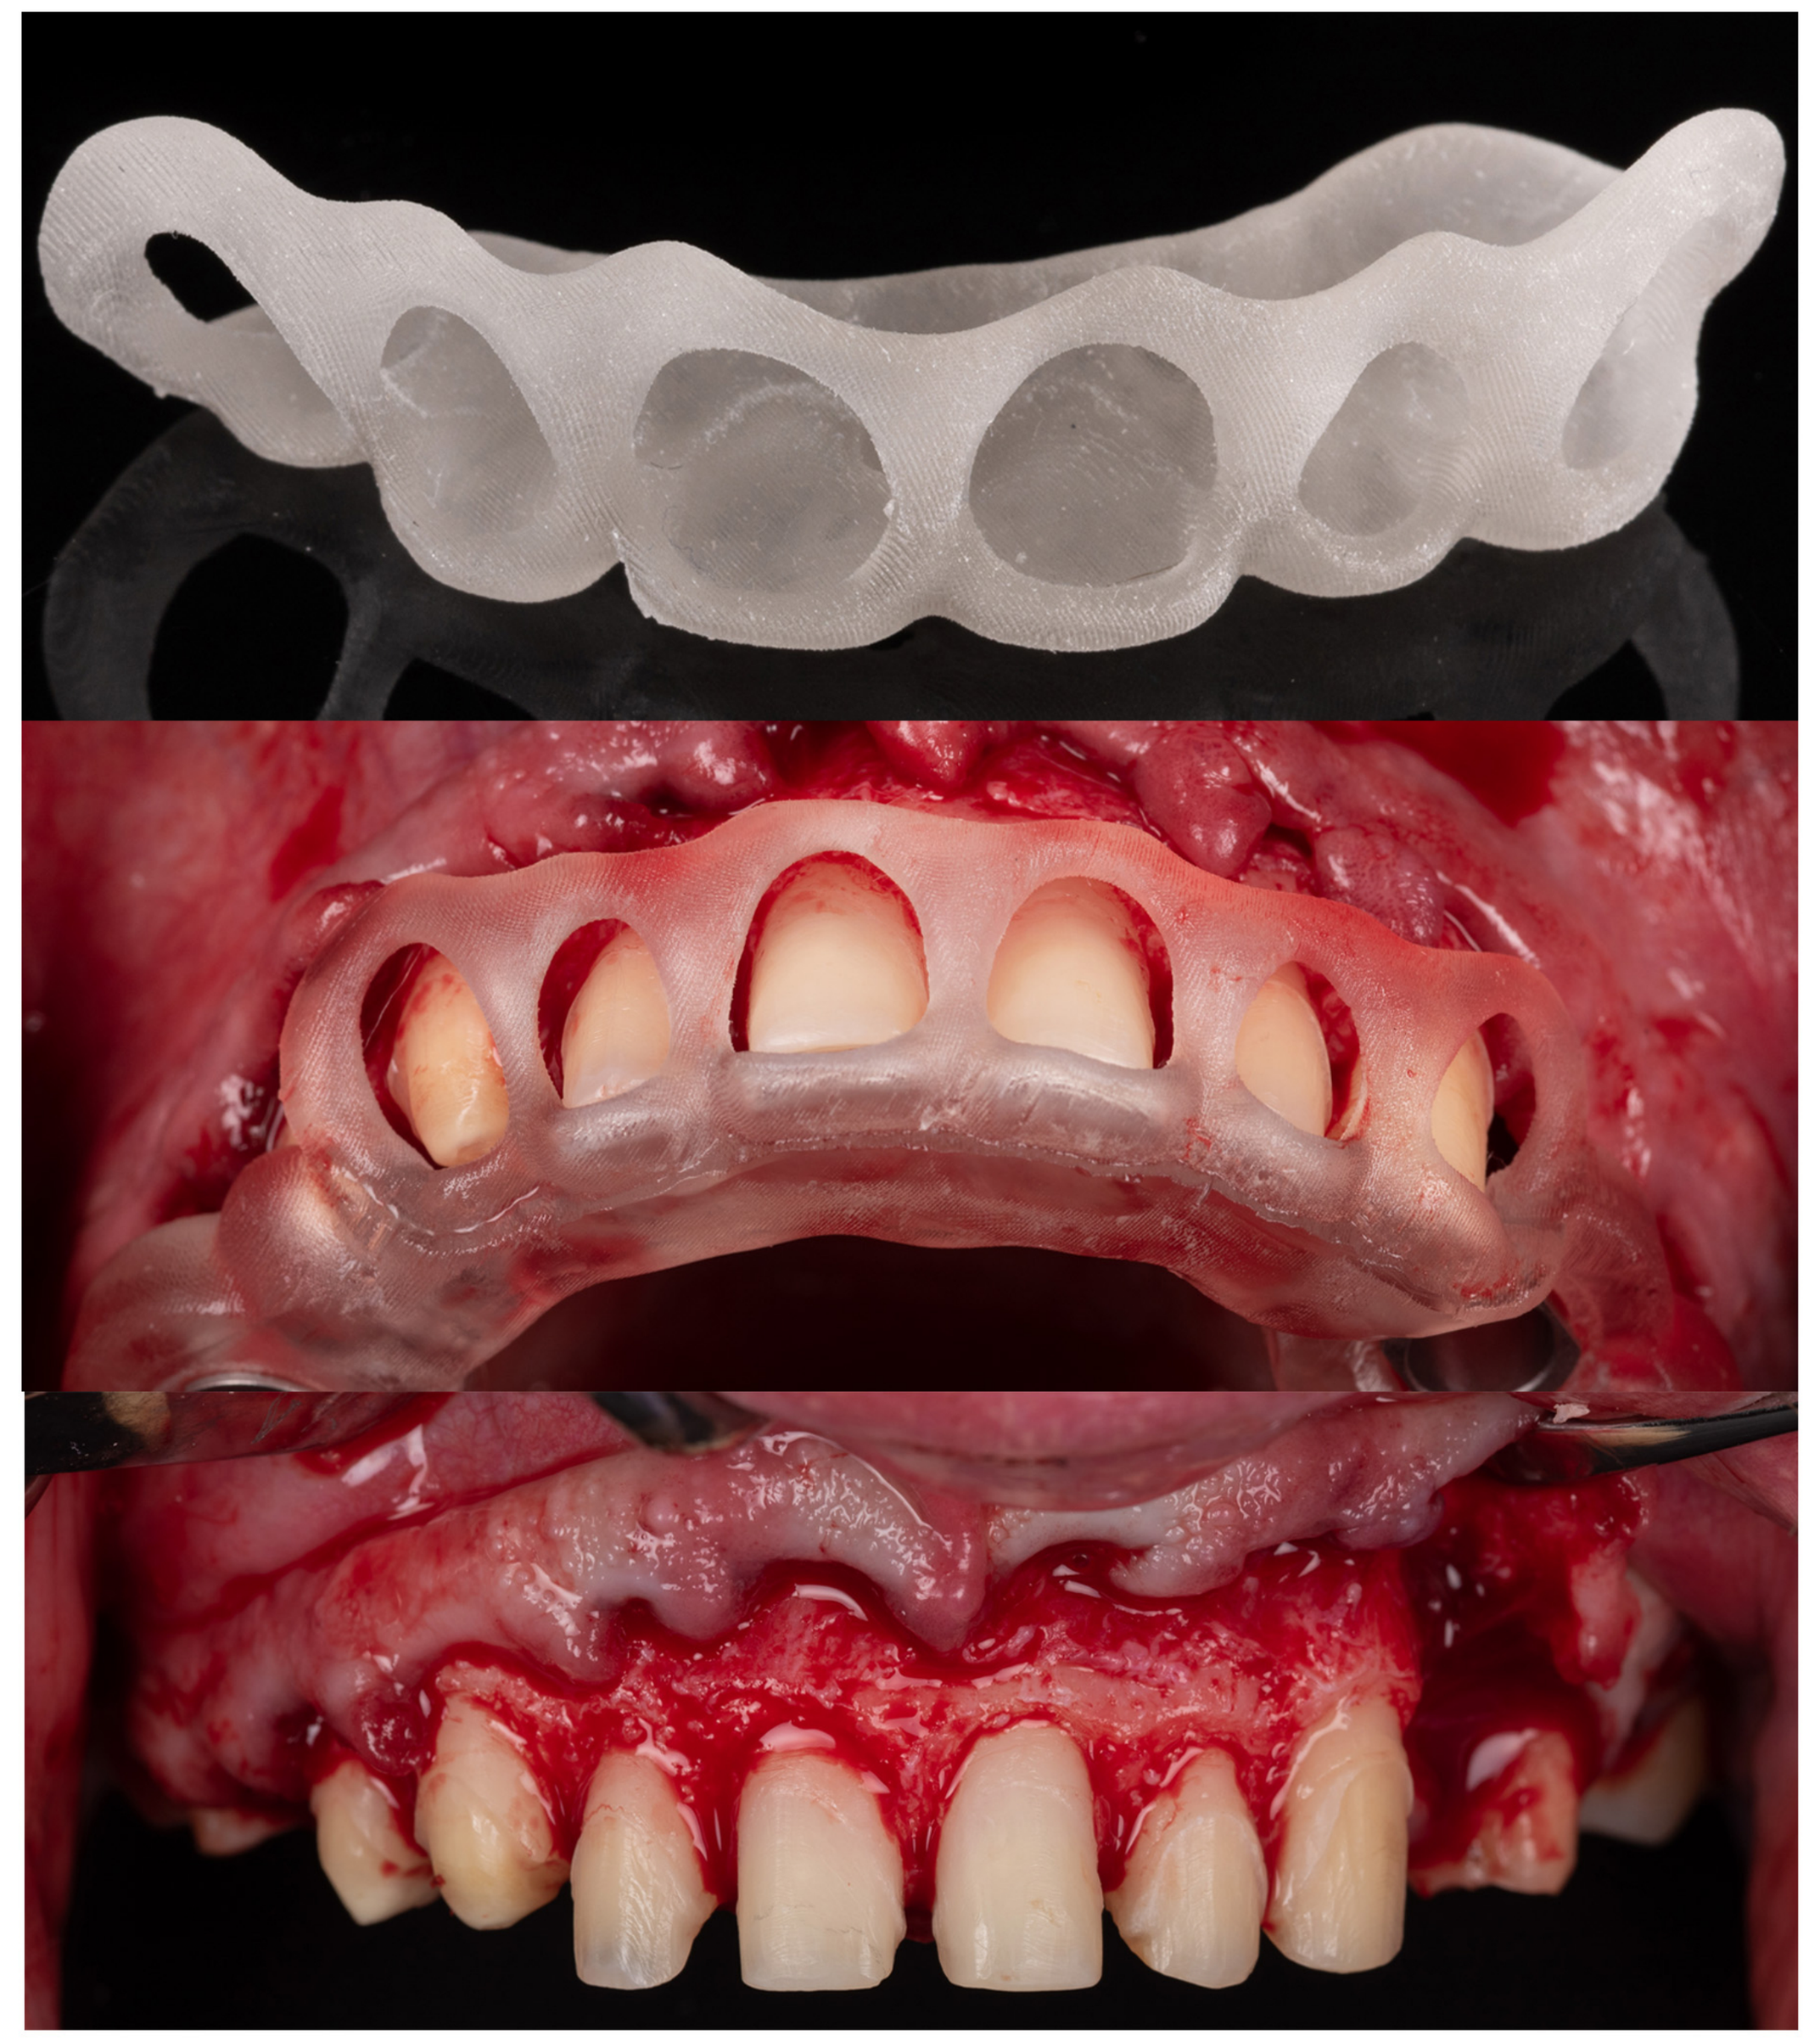

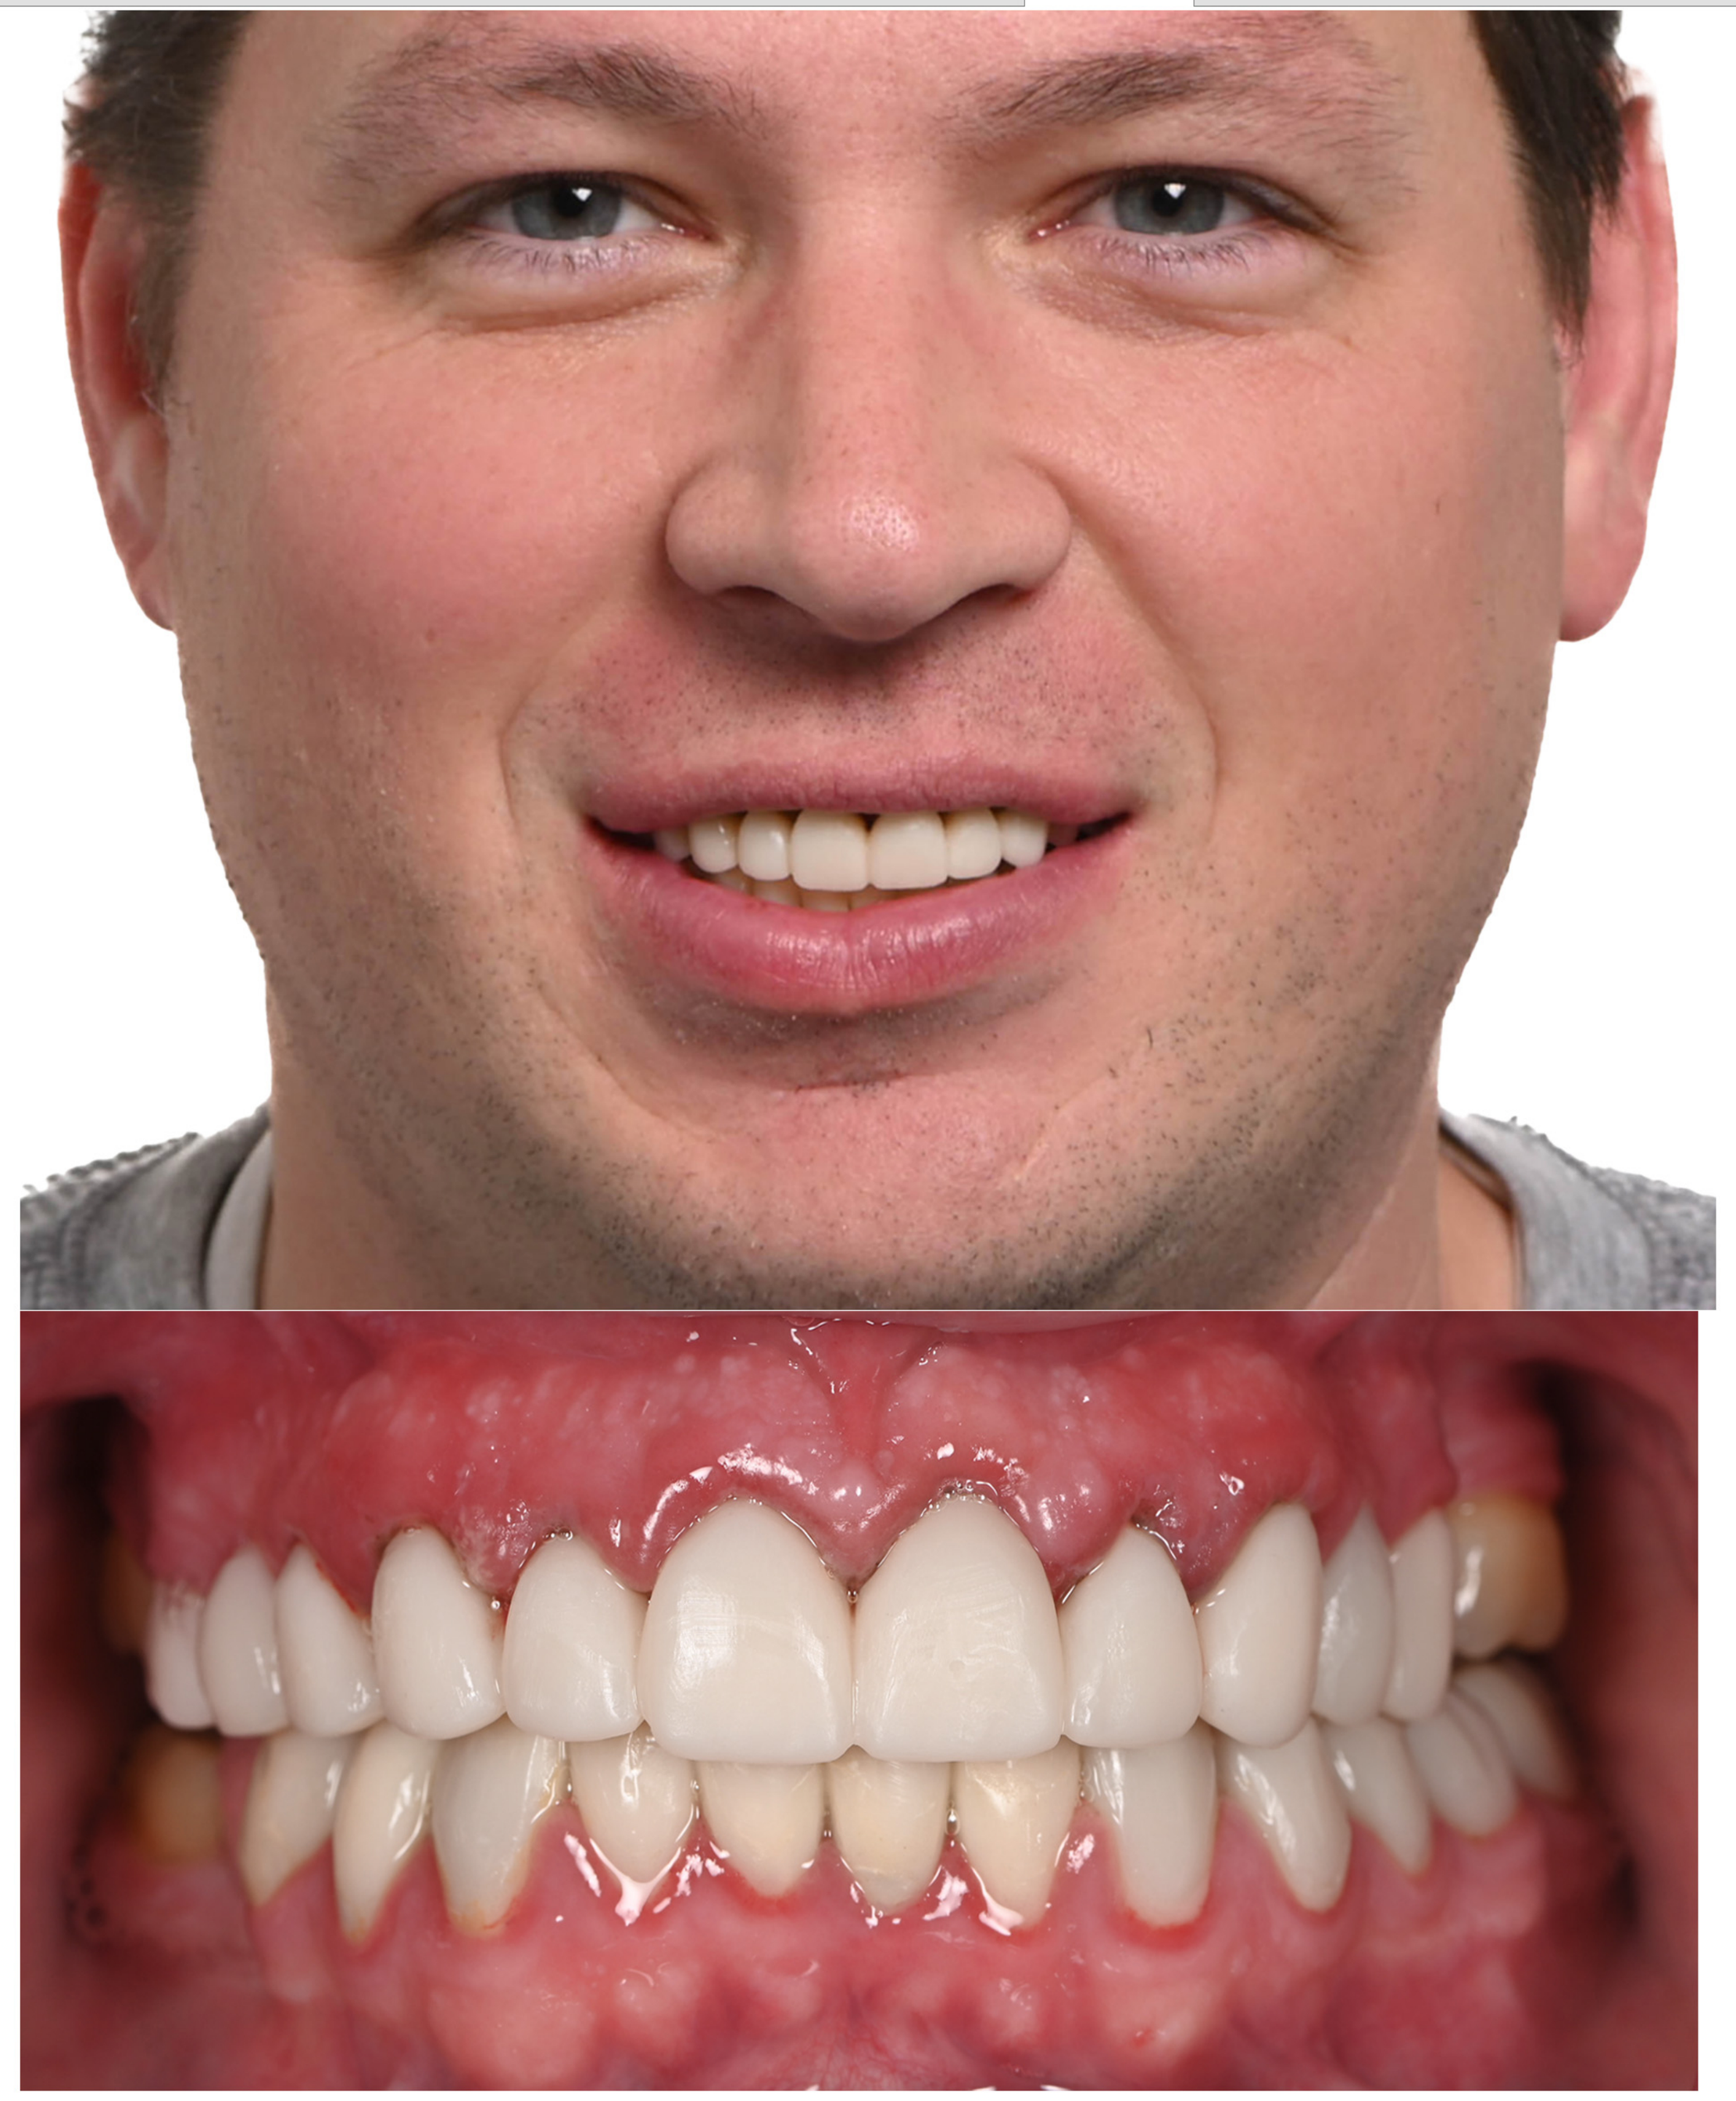

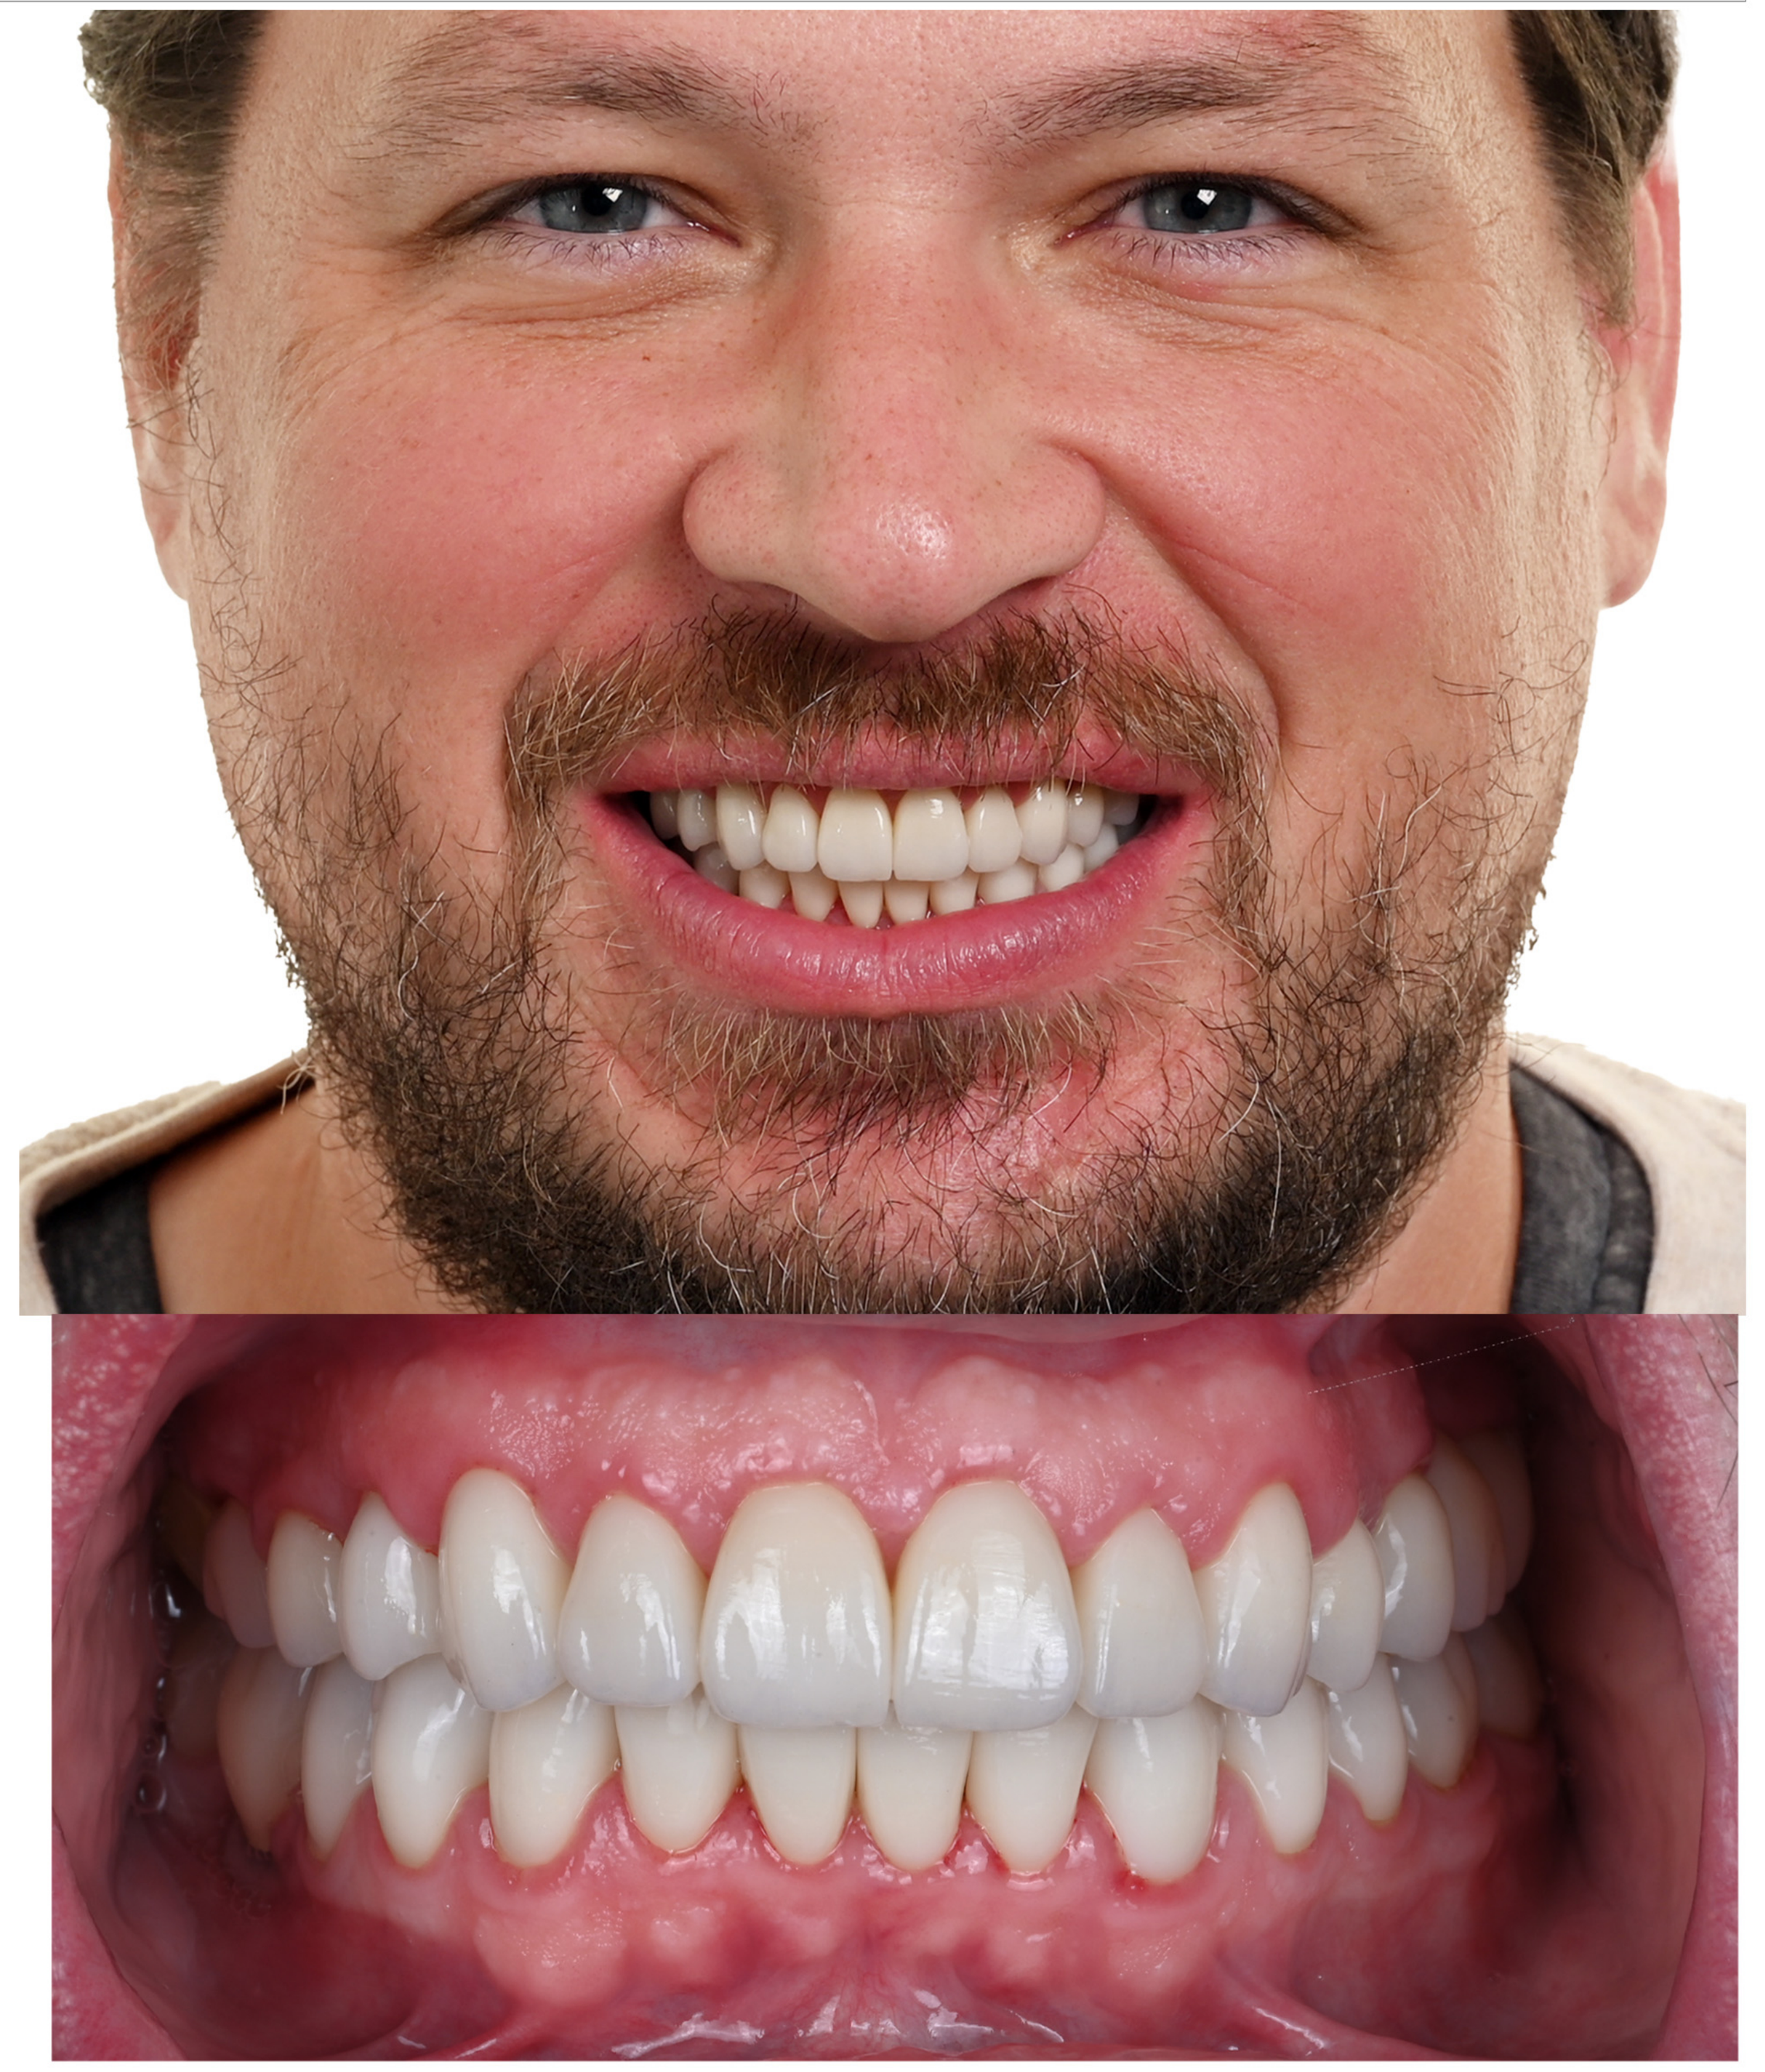