Investigating Same-Day Discharge For Postmastectomy Immediate Breast Reconstruction: A National Surgical Quality Improvement Program Study

Elijah G. Bingham, BS[1], Amanda Miller, BS[1], Nirbhay S. Jain, MD[1], Michael W. Wells, MEng[2], Andrew Da Lio, MD[1], Jason Roostaeian, MD[1], Christopher Crisera, MD[1], Ginger Slack, MD[1], Charles Tseng, MD[1], Michael Delong, MD[1], Jaco H. Festekjian, MD[1]

[1]David Geffen School of Medicine, University of California Los Angeles, Los Angeles, CA, USA, [2]Case Western Reserve University School of Medicine, Cleveland, OH, USA.

Introduction

- Patients undergoing immediate breast reconstruction with tissue expanders are frequently admitted postoperatively for monitoring and pain control,
- Need for aggressive pain control has been less necessary given the increased popularity of prepectoral reconstruction and the introduction of enhanced recovery protocols. Longer hospital stays also stress hospital resources and increase risks of nosocomial infection.
- If proven safe, same-day discharge could conserve resources, mitigate risk, and return patients home for faster recovery
- We used large data sets to investigate the safety of same-day discharge after mastectomy with immediate postoperative expander placement.

Methods

- We performed a retrospective review of patients in the National Surgical Quality Improvement Program (NSQIP) database who underwent tissue expander breast reconstruction between 2005 and 2019
- Patients were grouped based on date of discharge
- Demographic information, medical comorbidities, and outcomes were recorded.
- Statistical analysis was performed to determine efficacy of same-day discharge and identify factors predicting complication

Results

- Of the 14,387 included patients, 10% were discharged same-day, 70% postoperative day 1, and 20% later.
- Common complications such as infection, reoperation, and readmission trended upward with length of stay (6.4% v 9.3% v 16.8%), though were statistically equivalent between same-day and next-day discharge in both unilateral and bilateral surgeries.
- Patients discharged later had significantly more comorbidities than same or next-day discharge counterparts, and statistically higher complications rates.
- Predictors of complication included hypertension, smoking, diabetes, and obesity.

	Same Day Discharge *(n=1456)*	**POD1 Discharge** *(n=9377)*	**≥ POD2 Discharge** *(n=3554)*
All Complications	93 (6.4%)	873 (9.3%, $p < 0.001$)	596 (16.8%, $p < 0.001$)
Bleeding	4 (0.3%)	9 (0.1%, $p = 0.07$)	77 (2.2%, $p < 0.001$)
Infection	45 (3.1%)	340 (3.6%, $p = 0.3$)	164 (4.6%, $p = 0.01$)
Reoperation	52 (3.6%)	531 (5.7%, $p < 0.001$)	400 (11.2%, $p < 0.001$)
Readmission	30 (2.1%)	443 (4.7%, $p < 0.001$)	240 (6.7%, $p < 0.001$)

Table 1. Rates of various complications categorized by time of discharge.

Discussion

- We found that patients who were discharged the same day and postoperative day one had similar complication profiles. Only for patients with prolonged hospital courses were there increased risks of complications, readmission, and reoperation for patients.
- When controlling for preoperative comorbidities, earlier day of discharge did not predict an increased rate in complications.
- Same-day discharge adds a significant cost savings benefit to the hospital and patient. Based on publicly available information, the average cost of overnight surgery at large-scale academic centers is approximately $62,500 in hospital fees, which includes hoteling, nursing, medication, and other care, while the cost of same-day surgery is approximately $18,000 in hospital fees. Thus, same day surgery could save patients as much as $44,500.
- Future studies should include a multicenter prospective randomized controlled trial between same-day and next-day discharge.

Conclusions

- Same-day discharge after immediate tissue expander reconstruction has equivalent risk of perioperative complications as next-day discharge.
- For the otherwise healthy patient, going home the day of surgery is a safe option that minimizes costs and optimizes recovery while preserving postoperative outcomes.