Evaluation Of The Relative Safety Of Oncoplastic Reduction Compared To Traditional Reduction

Elijah Bingham, BS, Shamit Prabhu, MD, Nirbhay S. Jain, MD, Andrew Da Lio, MD, Charles Tseng, MD, Christopher Crisera, MD, Jaco Festekjian, MD, Jason Roostaeian, MD, Ginger Slack, MD, Michael Delong, MD.

Division of Plastic Surgery, University of California Los Angeles, Los Angeles, California, USA

Introduction

- Oncoplastic reduction (OnR) combines breast conservation therapy with reduction mammoplasty to provide effective tumor resection while achieving aesthetically pleasing surgical outcomes.
- Oncoplastic reduction theoretically includes the aggregate risks of both lumpectomy and bilateral breast reduction (BBR) surgeries
- Our goal was to understand if oncoplastic reduction confers a higher operative risk profile than bilateral breast reduction alone

Methods

- Patients from the National Surgical Quality Improvement Program (NSQIP) were identified by appropriate codes (19318 for BBR, 19301 or 19302 with 19318 for OBR)
- Demographics, preoperative comorbidities, and postoperative complications were extracted and analyzed with univariate and multivariate regressions.

Results

- 40,618 patients were included (BBR n=38,461, OBR n=2157)
- Compared to BBR patients, OBR patients were older (54.5 vs 41.1), more frequently Caucasian (63.3% vs 44.2%), with a higher BMI (32.4 vs 30.9), more medical comorbidities (10.9% DM, 37.2% HTN), and worse ASA (33.6% with score >2).
- OBR patients had higher rates of all complications (8.2% v 6%, p<0.001), reoperation (2.5% v 1.5%, p<0 .001), and readmission (2.5% v 1.3%, p <0.001).
- When controlling for confounding variables, OBR predicted higher reoperation and readmission at 30 days.

	BBR (n = 38461)	OnR (n = 2157)	p-value
PREOPERATIVE			
Age	41.4	54.5	< 0.001
BMI	30.9	32.4	< 0.001
Race (%)			< 0.001
White	44.3	63.3	
Asian	0.8	1.8	
Black	20.7	15.8	
Other	0.4	0.4	
Unreported	33.7	18.7	
DM (%)	4.5	10.9	< 0.001
Smoking (%)	8.8	7.3	0.018
Dyspnea (%)	1.7	3.8	< 0.001
Poor functional status (%)	0.5	0.6	0.366
COPD (%)	0.7	1.1	0.018
HTN (%)	18.1	37.2	< 0.001
Metastatic cancer (%)	0.1	0.6	< 0.001
Steroid use (%)	1.3	1.8	0.059
Bleeding disorder (%)	0.4	0.7	0.015
ASA (%)			< 0.001
1	27.0	8.1	
2	59.6	58.3	
Higher	13.4	33.6	

Table 1. Patient demographics and preoperative characteristics.

	BBR (n = 38461)	OnR (n = 2157)	p-value
POSTOPERATIVE			
Op Time	152	172	< 0.001
SSI			
(%)	3.4	4.0	0.369
POD	17	19	0.184
Dehiscence			
(%)	0.7	0.9	0.594
POD	17	18	0.876
Bleed			
(%)	0.2	0.3	0.490
POD	1	2	0.648
DVT/PE			
(%)	0.1	0.2	0.026
POD	9	15	0.074
Reoperation			
(%)	1.5	2.5	< 0.001
POD	8	13	0.119
Readmission			
(%)	1.3	2.5	< 0.001
POD	12	17	0.629
POD d/c	0	1	< 0.001

Table 2. Intraoperative variables and postoperative complications.

Discussion

- As the popularity of breast-conserving therapy continues to expand, oncoplastic breast reduction (OBR) serves as an extension of breast-conserving therapy to achieve better aesthetic outcomes and higher patient satisfaction scores for patients with larger tumors and larger breasts.
- After controlling for preoperative characteristics, OnR was not an independent predictor of total complications
- The presence of OBR, as compared to BBR, was only predictive of readmission and reoperation rates. However, the absolute increase in rate of readmission and reoperation is from approximately 1% to 2%, which is of unclear clinical significance.
- OnR has a slightly higher surgical risk profile than BBR but it is acceptable given the differing patient populations
- The discrepancy between racial groups suggests that access to OBR in underserved minority patients may also be lacking. This care gap deserves further investigation

Conclusions

- Patients undergoing OnR have higher rates of reoperation and readmission than BBR, likely due to the combination of greater comorbidities and additional surgical burden
- After controlling for preoperative characteristics, OnR was not an independent predictor of total complications
- OnR has a slightly higher surgical risk profile than BBR but it is acceptable given the differing patient populations