Sat-AIC-067
Mon-0660

Cross-Sectional Evaluation of Diffusion Tensor Imaging Endpoints Using Three Clinical Trials in Alzheimer's Disease

Diana Svaldi Otero,[1] Ilke Tunali,[1] Ajay Kurani,[1] David C. Airey,[1] Jiahe Li,[1] Isabel Wen Xu,[1] Jiaying Guo,[1] Hong Wang,[1] Emel SM Nery,[1] Emily C Collins,[1] Adam S Fleisher,[1] Kevin Biglan,[1] John R Sims,[1] Sergey Shcherbinin[1]

[1]Eli Lilly and Company, Indianapolis, USA

Sponsored by Eli Lilly and Company

Scan the QR code for a list of all Lilly content presented at the congress. Other company and product names are trademarks of their respective owners.

OBJECTIVE

- To perform exploratory, cross-sectional clinical validation of baseline free-water diffusion tensor imaging (FW-DTI) measures collected across three clinical trials in participants with early symptomatic Alzheimer's disease (AD)

CONCLUSION

- This comparative, exploratory analysis of FW-DTI endpoints across three clinical trials is consistent with prior evidence that multi-parametric FW-DTI may be used to capture disease-related microstructural changes in the brain that may complement volumetric MRI[3]
- A cohort with confirmed tau pathology and higher proportion of *APOE* ε4 carriers (PROSPECT-ALZ) demonstrated higher baseline extracellular and gray matter tissue pathology, as measured using FW-DTI, versus cohorts with only confirmed amyloid pathology
- FW-DTI measures showed more similarity across populations with equivalent enrollment criteria (TRAILRUNNER-ALZ 1 and TRAILBLAZER-ALZ 6), reflecting similar clinical and pathological levels of disease
- These findings may suggest that FW-DTI can differentiate levels of neurodegeneration in clinical research settings

Alzheimer's Association International Conference 2026; London, United Kingdom, and Online; July 12–15, 2026

BACKGROUND

- Progression of AD is associated with microstructural changes in the brain that often precede atrophy[1]
- FW-DTI is an MRI technique that characterizes the diffusion of water molecules in tissue[2] and can be used to measure microstructural changes in the brain[3]
- Baseline diffusion scans were available from sub-studies of three late-phase, multicenter clinical trials in participants with early symptomatic AD with confirmed amyloid or tau pathology: TRAILRUNNER-ALZ 1 (NCT05463731),[4] TRAILBLAZER-ALZ 6 (NCT05738486),[5,6] and PROSPECT-ALZ (NCT05063539)[7,8]

METHODS

- Baseline diffusion scans from participants with available DTI data across three trials were analyzed
- Extracellular FW and tissue-specific diffusion properties were modeled using FW-DTI; measures by brain region are summarized in **Table 1** and **Figure 1**

Table 1: Measures Used in Different Brain Regions

Measure	Brain Regions Analyzed
Free-water (FW): Biomarker of extracellular diffusion (candidate marker of inflammation)	Hippocampus, whole cortex, cerebral white matter
Mean diffusivity (MD_T): Biomarker of gray-matter integrity	Hippocampus, whole cortex
Fractional anisotropy (FA_T): Biomarker of axonal integrity	Cerebral white matter

Figure 1: Variables Investigated

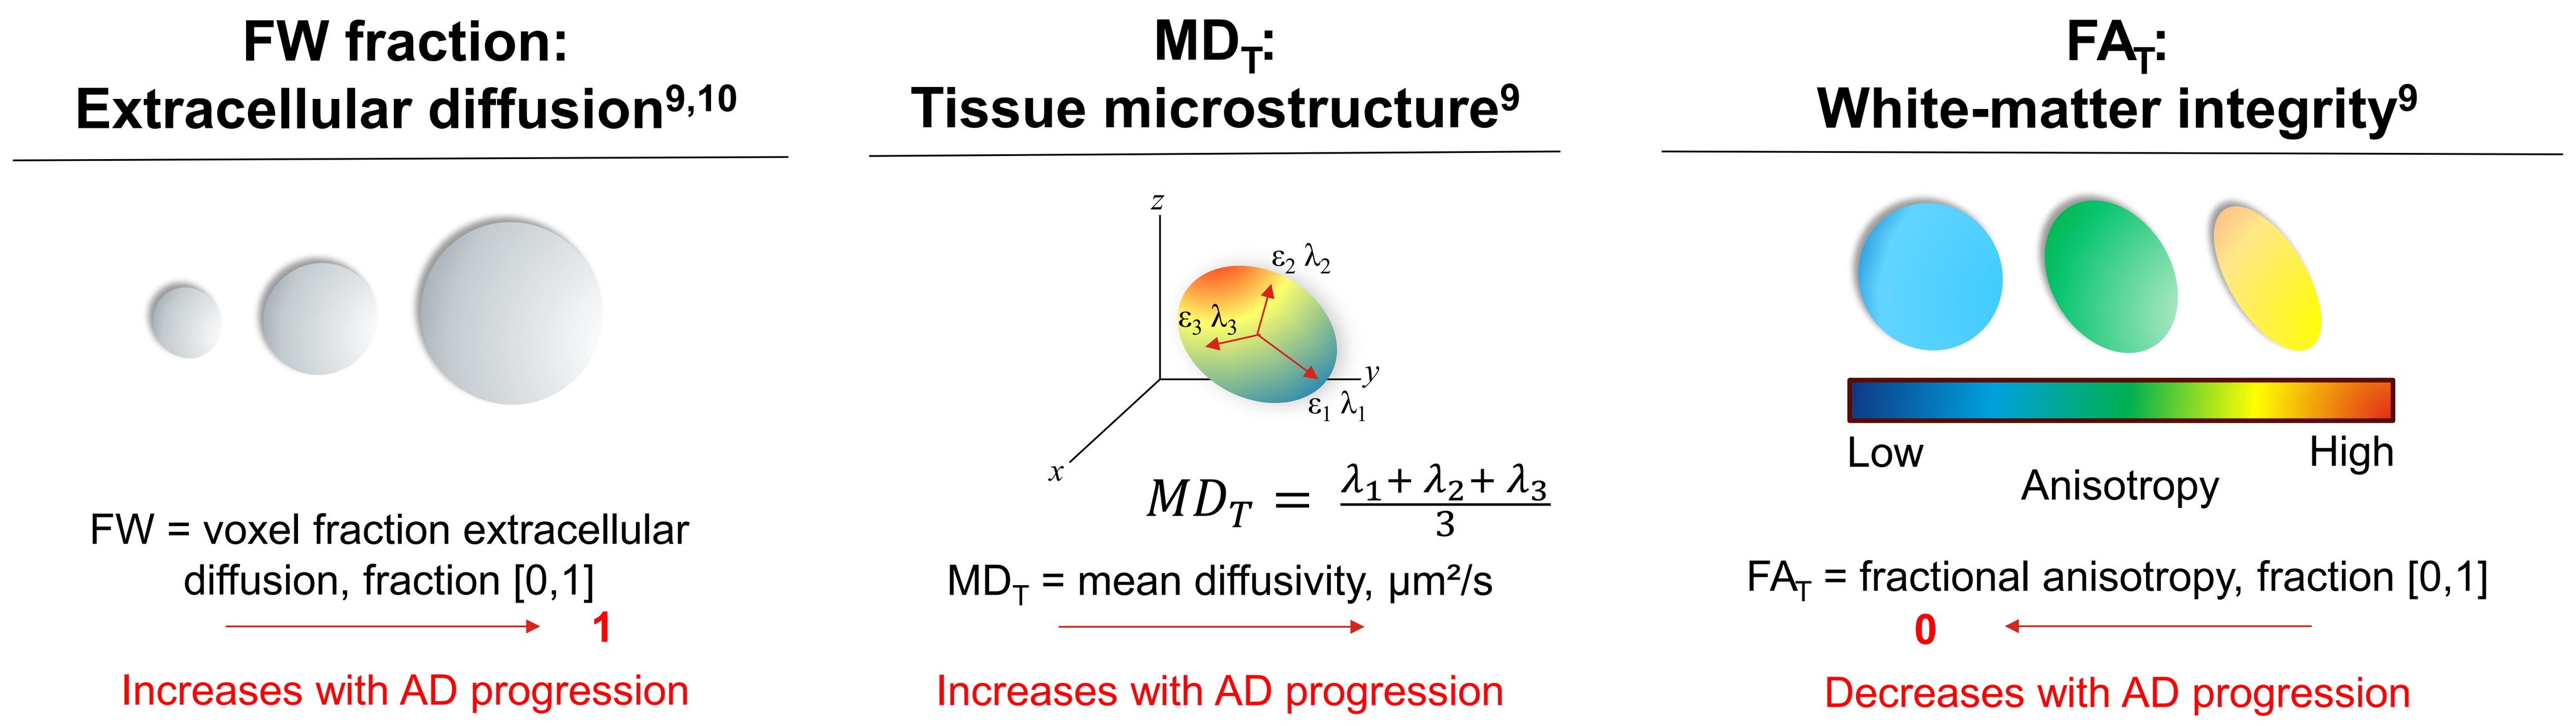
FW fraction:
Extracellular diffusion[9,10]
FW = voxel fraction extracellular diffusion, fraction [0,1]
1
Increases with AD progression
MD_T:
Tissue microstructure[9]
z
y
x
$\varepsilon_1\ \lambda_1$
$\varepsilon_2\ \lambda_2$
$\varepsilon_3\ \lambda_3$
$MD_T = \frac{\lambda_1 + \lambda_2 + \lambda_3}{3}$
MD_T = mean diffusivity, µm²/s
Increases with AD progression
FA_T:
White-matter integrity[9]
Low
Anisotropy
High
FA_T = fractional anisotropy, fraction [0,1]
0
Decreases with AD progression

RESULTS

Table 2: Key Baseline Characteristics and Biomarker Eligibility of Participants Analyzed Across Three Clinical Trials

Characteristic	Overall	TRAILRUNNER-ALZ 1	TRAILBLAZER-ALZ 6	PROSPECT-ALZ	*P* Value[a]
Participants, n	1,579	586	825	168	
Key baseline characteristics					
Mean age (SD), years	74 (6)	74 (6)	74 (6)	74 (5)	0.137
***APOE* ε4 carrier status**					**0.015**
Carriers, n (%)	1008 (64)	353 (60)	535 (65)	120 (72)	
Non-carriers, n (%)	569 (36)	233 (40)	289 (35)	47 (28)	
Mean (SD) MMSE Score	24.74 (2.57)	24.88 (2.58)	24.73 (2.49)	24.31 (2.80)	**0.040**
Biomarker eligibility criteria					
Amyloid pathology		Confirmed	Confirmed	Not measured	
Tau PET status		Not measured	Not measured	Low/medium to high tau levels[11]	
P-tau217 positivity		Not confirmed	Not confirmed	Confirmed	

[a]Indicates study differences in a given covariate, using analysis of variance test for continuous variables or Chi-square test for categorical variables.

Figure 2: FW, MD_T, and FA_T in Gray and White Matter[a] Were Higher in PROSPECT-ALZ Compared to Other Late-Phase Clinical Trials in Early Symptomatic AD

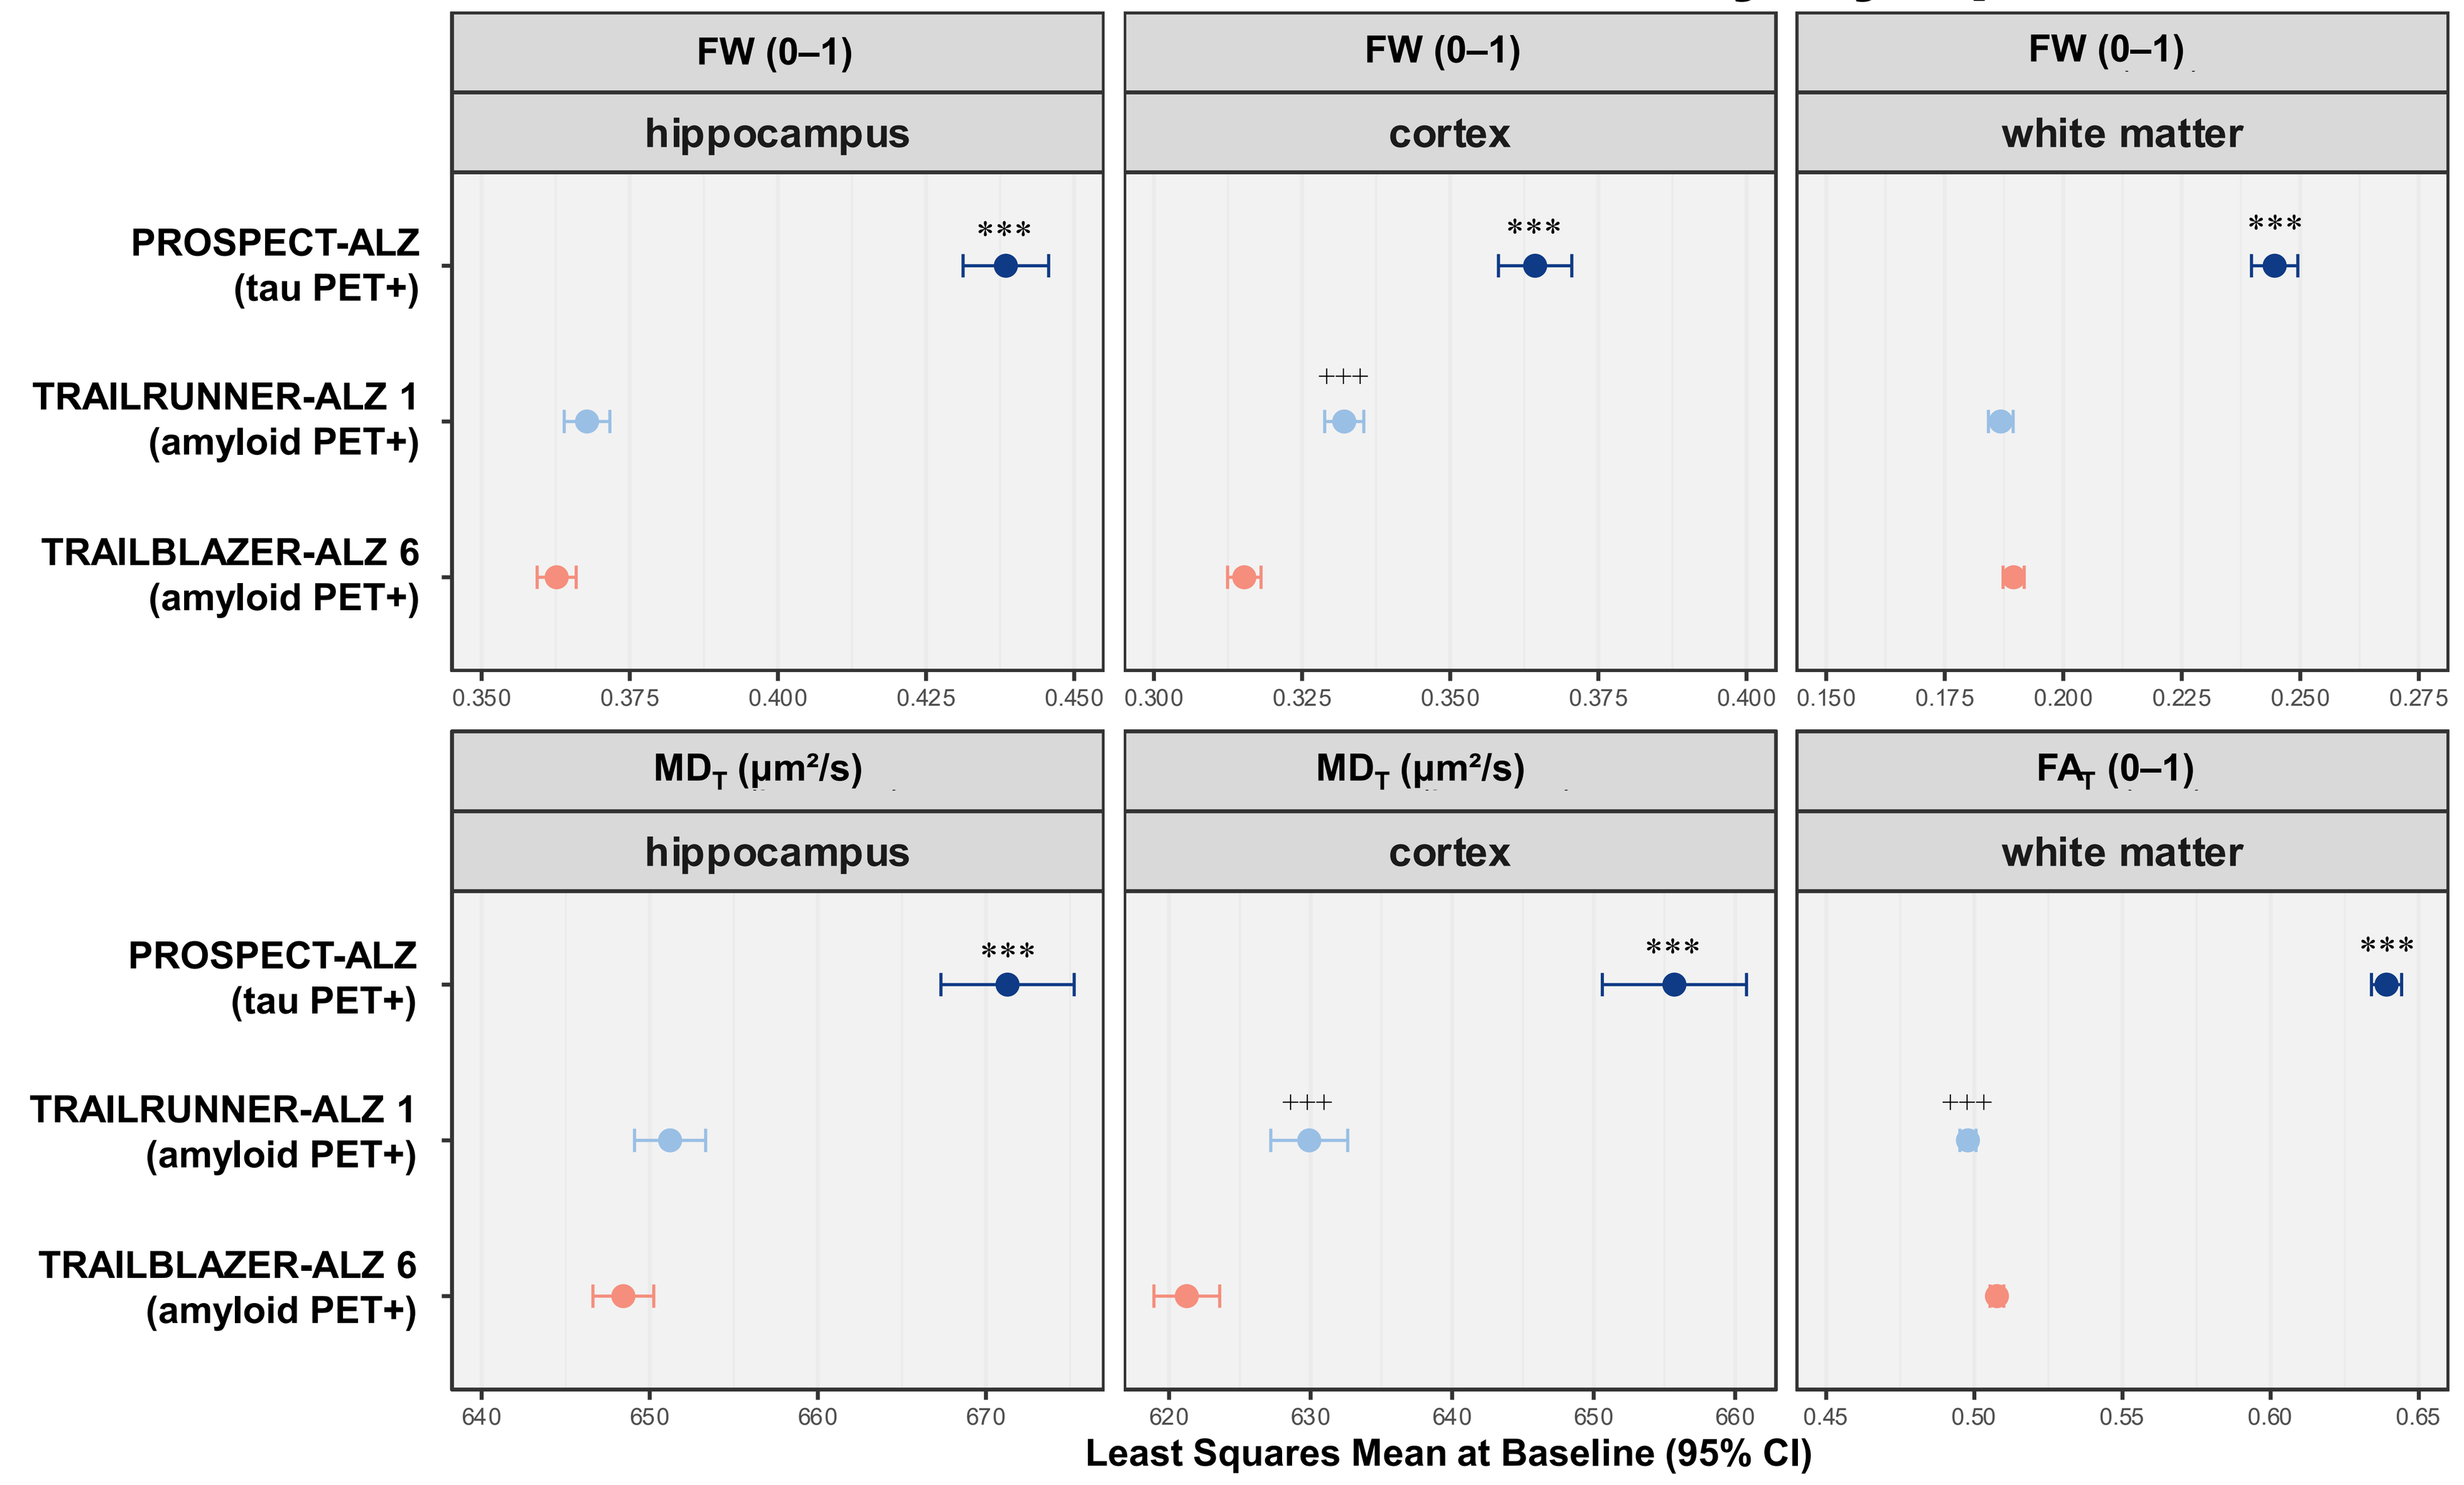
FW (0–1)
hippocampus
cortex
white matter
MD_T (µm²/s)
FA_T (0–1)
PROSPECT-ALZ (tau PET+)
TRAILRUNNER-ALZ 1 (amyloid PET+)
TRAILBLAZER-ALZ 6 (amyloid PET+)

+++
0.350 0.375 0.400 0.425 0.450
0.300 0.325 0.350 0.375 0.400
0.150 0.175 0.200 0.225 0.250 0.275
640 650 660 670
620 630 640 650 660
0.45 0.50 0.55 0.60 0.65
Least Squares Mean at Baseline (95% CI)

[a] Adjusted for baseline age, MMSE score, and *APOE* ε4 carrier status; Tukey-adjusted CIs; ***Significantly different from both TRAILRUNNER-ALZ 1 and TRAILBLAZER-ALZ 6; +++Significantly different from TRAILBLAZER-ALZ-6

- PROSPECT-ALZ had a higher proportion of *APOE* ε4 carriers (**Table 2**) among similar age ranges, suggesting that confirming eligibility by P-tau217 and tau PET positivity versus amyloid PET positivity may enrich for *APOE* ε4 carriers
- Least squares mean and 95% CI (adjusted for *APOE* ε4 carrier status and MMSE score) of each FW-DTI measure across clinical trials are shown in **Figure 2**
 - FW and MD_T estimates were numerically higher in the hippocampus versus cortex, consistent with the typical spatial spread of AD pathology
 - Hippocampal and white-matter FW and hippocampal MD_T were not significantly different between trials requiring only amyloid presence (TRAILRUNNER-ALZ 1 and TRAILBLAZER-ALZ 6) but were higher in PROSPECT-ALZ, which required tau positivity
 - The pairwise differences in magnitude were larger (10× greater cortical MD_T, 2× greater white-matter FA_T) for PROSPECT-ALZ versus other trials than for TRAILBLAZER-ALZ 6 versus TRAILRUNNER-ALZ 1

Abbreviations: AD, Alzheimer's disease; *APOE* ε4, apolipoprotein E ε4 allele; CI, confidence interval; DTI, diffusion tensor imaging; FA_T, fractional anisotropy, FW, free-water; FW-DTI, free-water DTI; MD_T, mean diffusivity; MMSE, mini mental state examination; MRI, magnetic resonance imaging; P-tau217, phosphorylated tau217; PET, positron emission tomography; SD, standard deviation

References:
1. Debatisse J, et al. *Cells*. 2025;14:155.
2. Tae WS, et al. *J Clin Neurol*. 2018;14:129-140.
3. Bergamino M, et al. *Sci Rep*. 2021;11:6990.
4. Hufford M, et al. Presented at CTAD 2025. Poster P-040.
5. Wang H, et al. *J Prev Alzheimers Dis*. 2025;12:100266.
6. Wang H, et al. *Alzheimers Dement*. 2025;21:e70062.
7. Munsie LM, et al. *Alzheimers Dement*. 2023;19(Suppl 21):e074725.
8. Munsie LM, et al. Presented at: CTAD 2025. Late Breaking Communication LB5.
9. Pasternak O, et al. *Magn Reson Med*. 2009;62:717-730.
10. Macey PM, et al. *Heliyon*. 2018;4:e00700.
11. Pontecorvo MJ, et al. *Brain*. 2019;142:1723–1735.

Disclosures: All authors are full-time employees and shareholders of Eli Lilly and Company.
Acknowledgments: Medical writing support was provided by Scott Salsman, PhD, CMPP, and editorial support was provided by Aruna Colaco Clemente of Syneos Health. funded by Eli Lilly and Company.

Copyright ©2026 Eli Lilly and Company. All rights reserved.