Poster Code: aaic2026.a3612bc
Poster Number: Monday-0225

MRI–pathology correlation of cerebral amyloid angiopathy focusing on small vessel disease scores

Akihiro Shindo, Hidehiro Ishikawa, Shotaro Horie, Takaya Utsunomiya, Hirofumi Matsuyama, Atsushi Niwa

Department of Neurology, Graduate School of Medicine, Mie University, Japan

Background: With the advent of anti-amyloid-β antibody therapies, the management of Alzheimer's disease has entered a new era. However, amyloid-related imaging abnormalities (ARIA) have emerged as an important adverse effect. Cerebral amyloid angiopathy (CAA) is considered a key risk factor for ARIA. Although patients with advanced CAA are excluded from anti-amyloid-β therapy, mild or subclinical CAA may still be present. Therefore, accurate MRI-based assessment of underlying CAA pathology is crucial in clinical practice.

Objective: To investigate the association between pathological severity of CAA and premortem MRI markers of cerebral small vessel disease (SVD), with particular focus on composite SVD scores.

Methods: We retrospectively analyzed 13 autopsy cases examined at our institution between 2008 and 2024. Cases with pathologically confirmed CAA (Vonsattel grade 1–2) and cases without CAA (Vonsattel grade 0) were included. Premortem brain MRI scans were reviewed for SVD markers, including lacunes, cerebral microbleeds, enlarged perivascular spaces, and white matter hyperintensities (WMH). WMH was defined using established Fazekas-based criteria, and Total SVD score, CAA-SVD score, and modified CAA-SVD score were calculated. In addition, multifocal WMH spot lesions were assessed. Correlations between imaging findings, SVD scores, and Vonsattel grade were evaluated using Spearman's rank correlation coefficient.

Results: Among the 13 cases, Vonsattel grade was 0 in 3 cases, 1 in 5 cases, and 2 in 5 cases. Vonsattel grade showed a moderate positive correlation with WMH burden (Spearman's ρ = 0.62, p = 0.025). Positive correlations were also observed between Vonsattel grade and the CAA-SVD score as well as the modified CAA-SVD score, although these associations were weaker than that observed for WMH burden. In contrast, hemorrhagic MRI markers, including lobar cerebral microbleeds and cortical superficial siderosis, demonstrated weak or no correlation with pathological CAA severity. The Total SVD score showed only a weak to moderate association with Vonsattel grade.

Conclusion: Pathological severity of cerebral amyloid angiopathy was more closely associated with white matter hyperintensity burden than with hemorrhagic MRI markers. These findings suggest that the presence of moderate to severe WMH burden on baseline MRI may indicate underlying CAA, even in the absence of prominent hemorrhagic lesions. In patients being considered for anti-amyloid-β antibody therapy, careful attention to WMH burden may therefore be important for MRI-based risk assessment of ARIA. Although the sample size was limited, this autopsy-based MRI–pathology correlation provides direct insight into imaging correlates of CAA severity.

INTRODUCTION

Anti-amyloid-β antibody therapies have transformed the management of early Alzheimer's disease but have also highlighted the clinical importance of ARIA.

Accurate MRI-based assessment of underlying CAA is therefore essential for ARIA risk assessment.

Why do we need better MRI markers of underlying CAA?

Baseline MRI (before lecanemab)
•Lobar cerebral microbleeds (CMBs) n = 2
•Enlarged centrum semiovale perivascular spaces (EPVS)
•**CAA-SVD score = 2**

•**Developed ARIA-E**

Representative clinical case

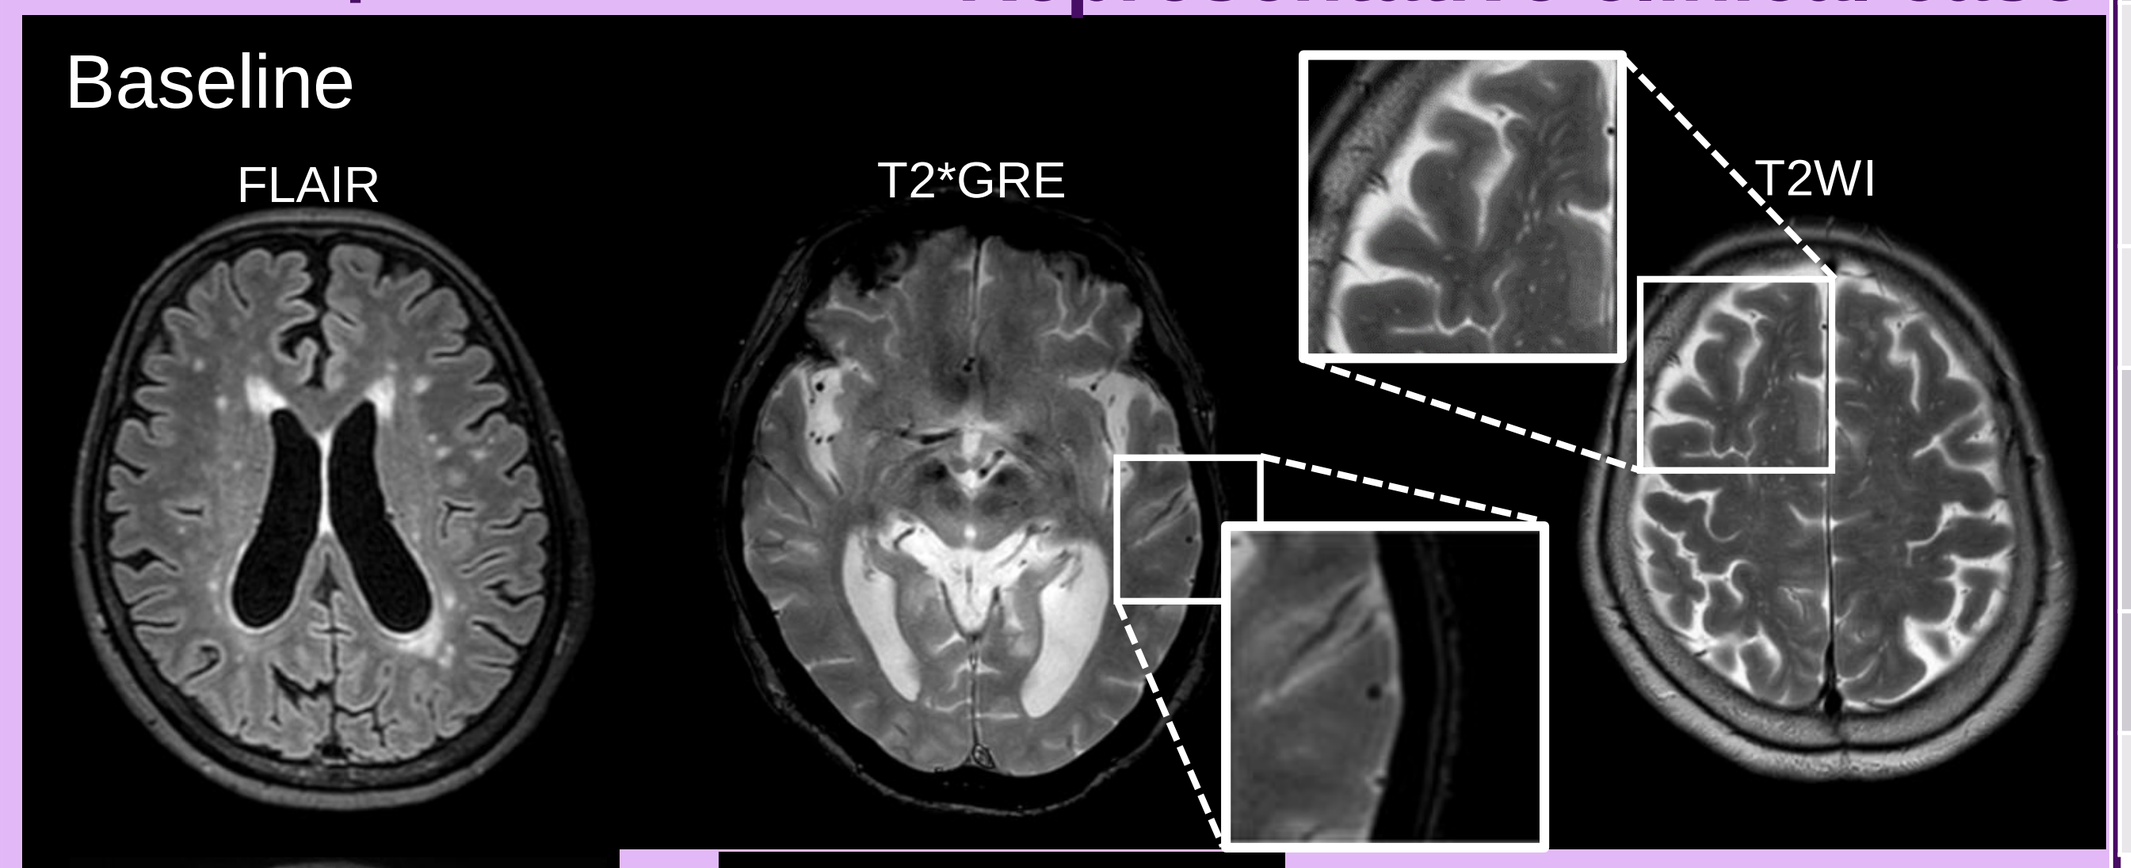
Baseline
FLAIR
T2*GRE
T2WI

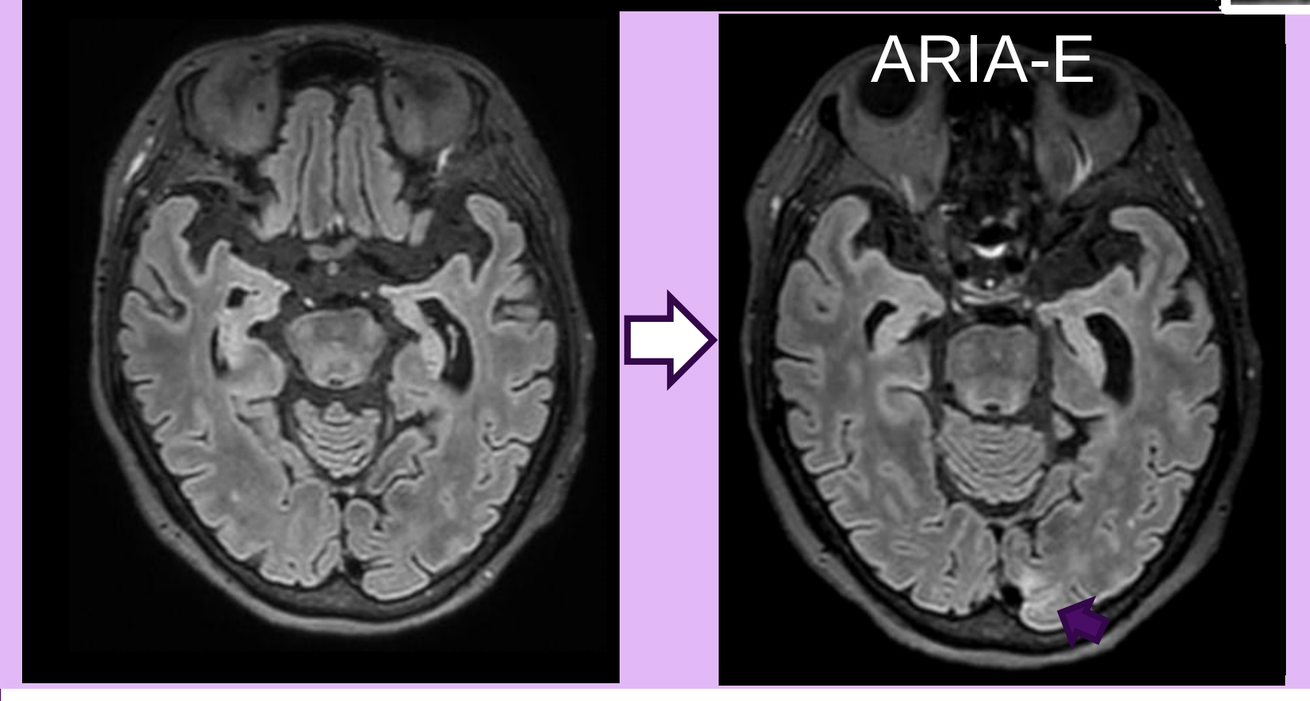
ARIA-E

What MRI findings reflect underlying pathological CAA?

Because advanced CAA is already excluded from treatment, identifying MRI markers of mild pathological CAA is crucial for improving ARIA risk assessment.

METHODS

■ Study population

Autopsy cases 2008-2024 n= 13
CAA Vonsattel grade 0–2

■ MRI assessment

WMH (Fazekas, multi spots)
Lobar CMBs, EPVS, Lacunes

■ MRI markers included in each SVD

	MRI marker	Total	CAA-SVD	modified
CMBs	Any	✓	–	–
CMBs	Lobar	–	✓	✓
	Cortical superficial siderosis	–	✓	✓
EPVS	Basal ganglia	✓	–	–
EPVS	Centrum semiovale		✓	✓
	Lacunes	✓	–	–
WMH	Deep and/or Periventricular	✓	✓	✓
WMH	Posterior Distribution	–	–	✓
	Cortical microinfarcts	–	–	✓
	Maximum score	4	6	8

Total SVD, CAA-SVD, and modified CAA-SVD scores were calculated. WMHs in a multispot pattern were also assessed.

■ Statistical analysis

Correlations between MRI findings and pathological CAA severity were evaluated using Spearman's rank correlation coefficient.

RESULTS

Association between Vonsattel grade and composite MRI SVD scores

SVD score	Spearman's ρ	p
Total SVD	0.29	0.34
CAA-SVD	0.34	0.26
modified CAA-SVD	0.28	0.36

No significant correlations were observed between Vonsattel grade and composite MRI SVD scores, including Total SVD, CAA-SVD, and modified CAA-SVD.

Association between individual MRI markers and Vonsattel grade

MRI marker	Spearman's ρ	p
Any CMB	-0.319	0.339
Lobar CMB	0.000	1.000
cSS	NA	NA
Basal ganglia EPVS	-0.082	0.789
Centrum semiovale EPVS	-0.143	0.642
Lacunes	-0.122	0.692
Deep and/or Periventricular WMH	**0.616**	**0.025**
Posterior distribution WMH	0.104	0.735
Cortical microinfarcts	-0.082	0.789

Among individual MRI markers, only WMH was significantly associated with pathological CAA severity (Vonsattel grade).

White matter hyperintensity burden across Vonsattel grades

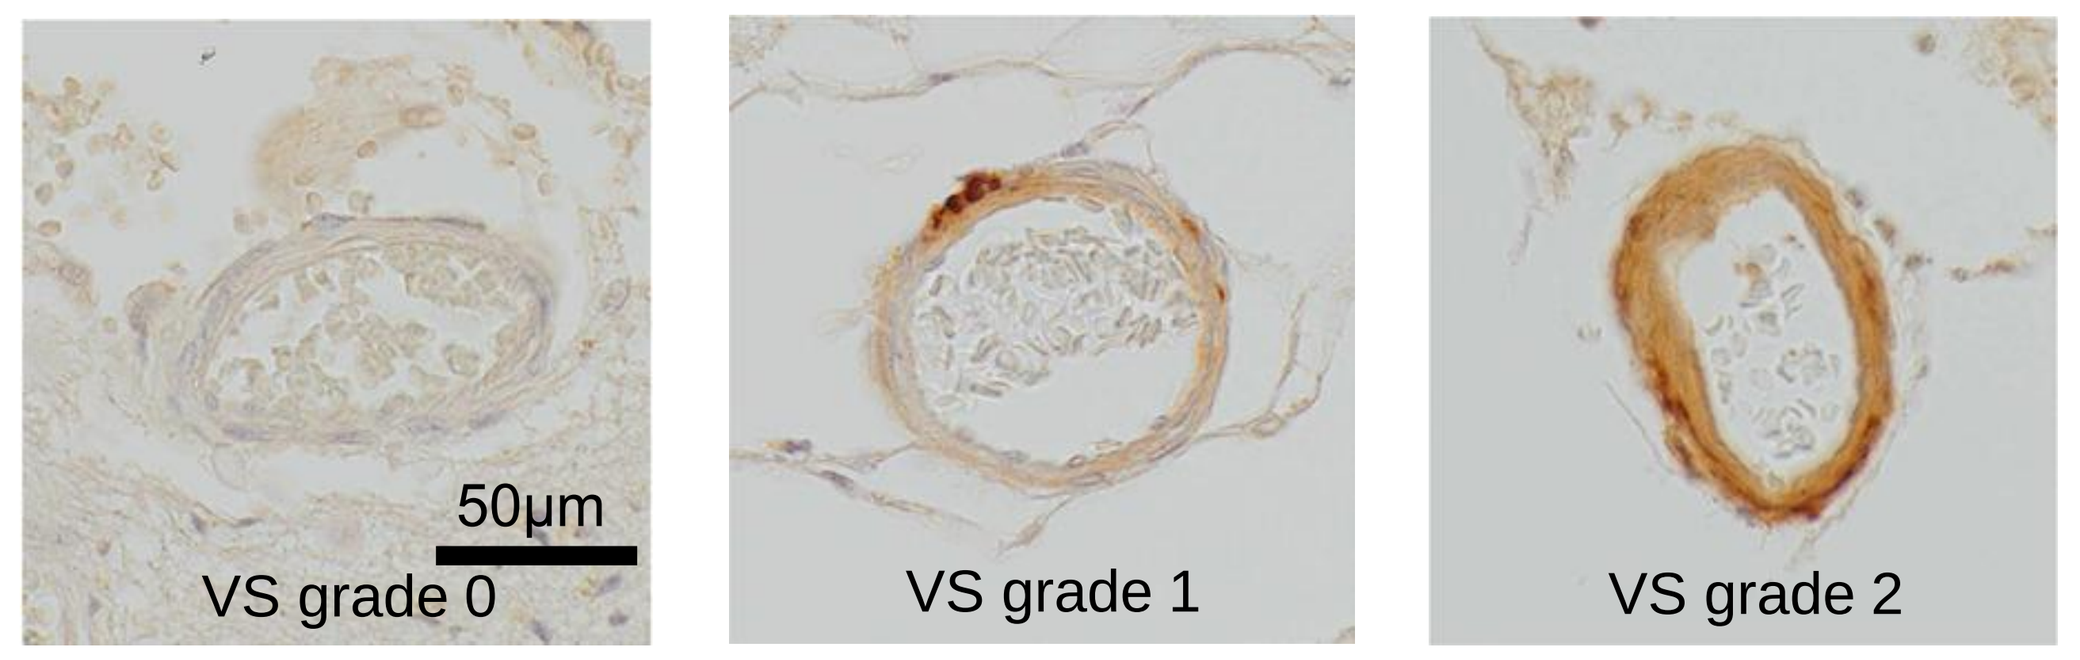
50μm
VS grade 0
VS grade 1
VS grade 2

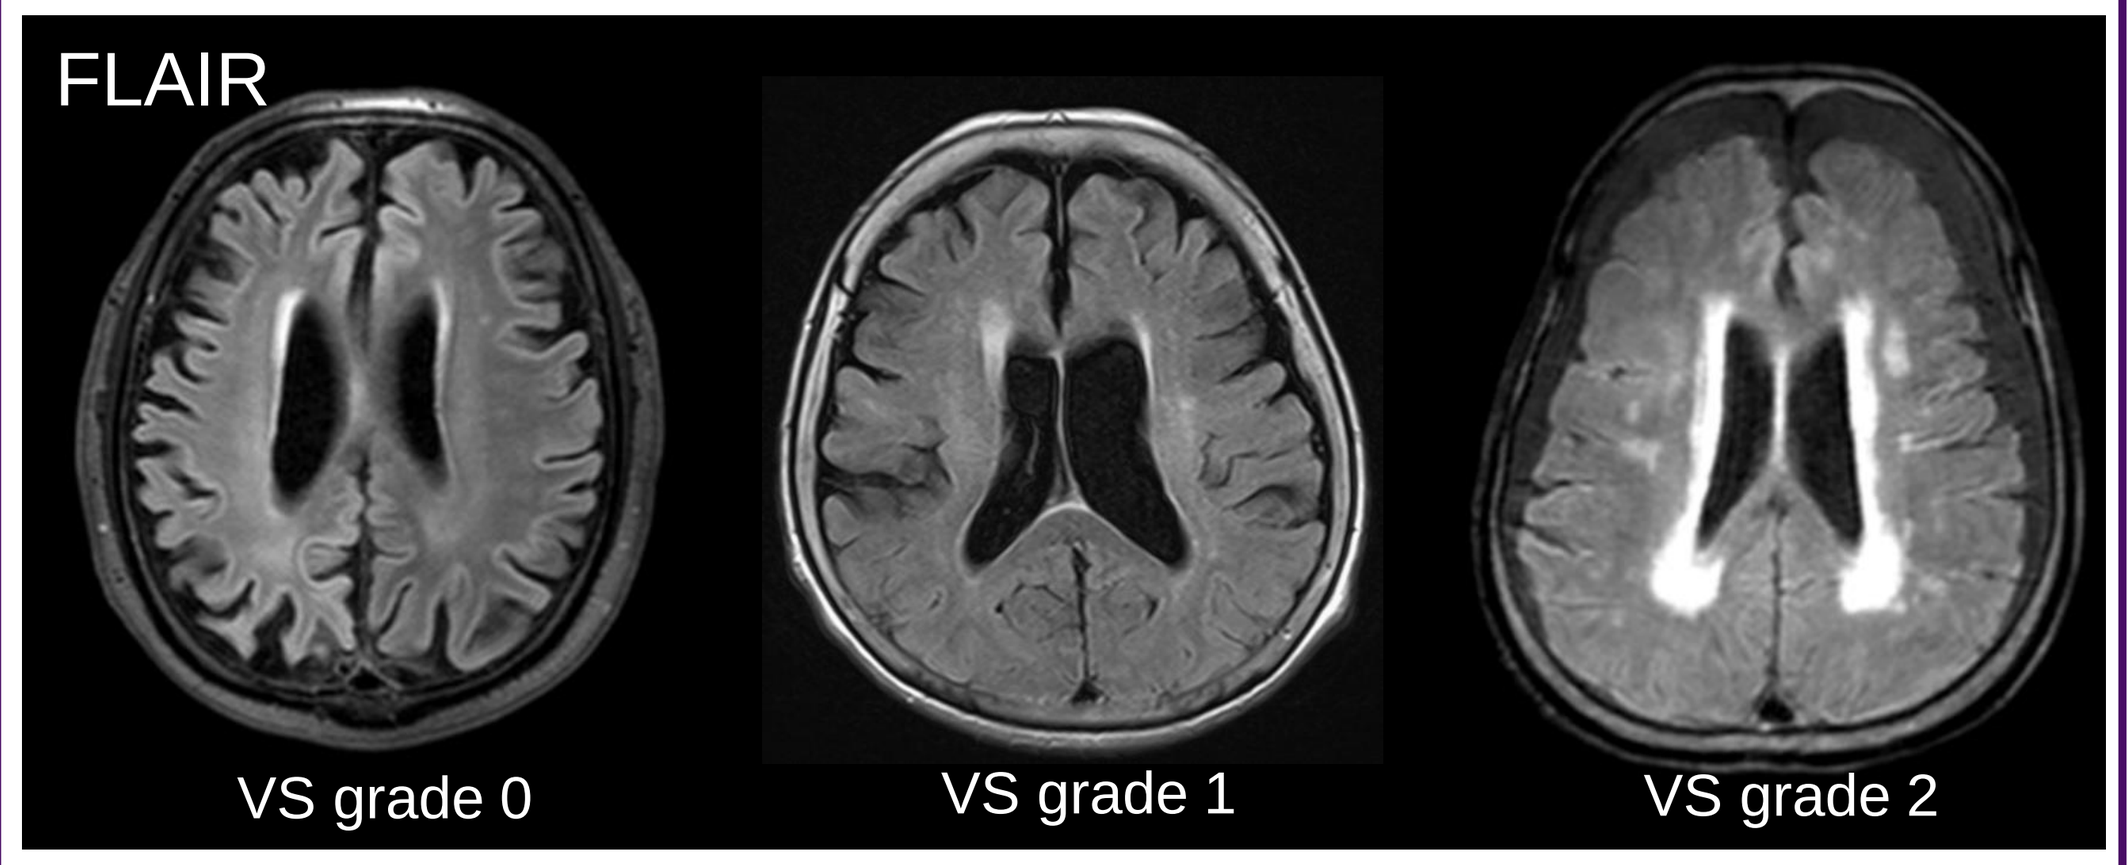
FLAIR
VS grade 0
VS grade 1
VS grade 2

Association between WMH severity and Vonsattel grade

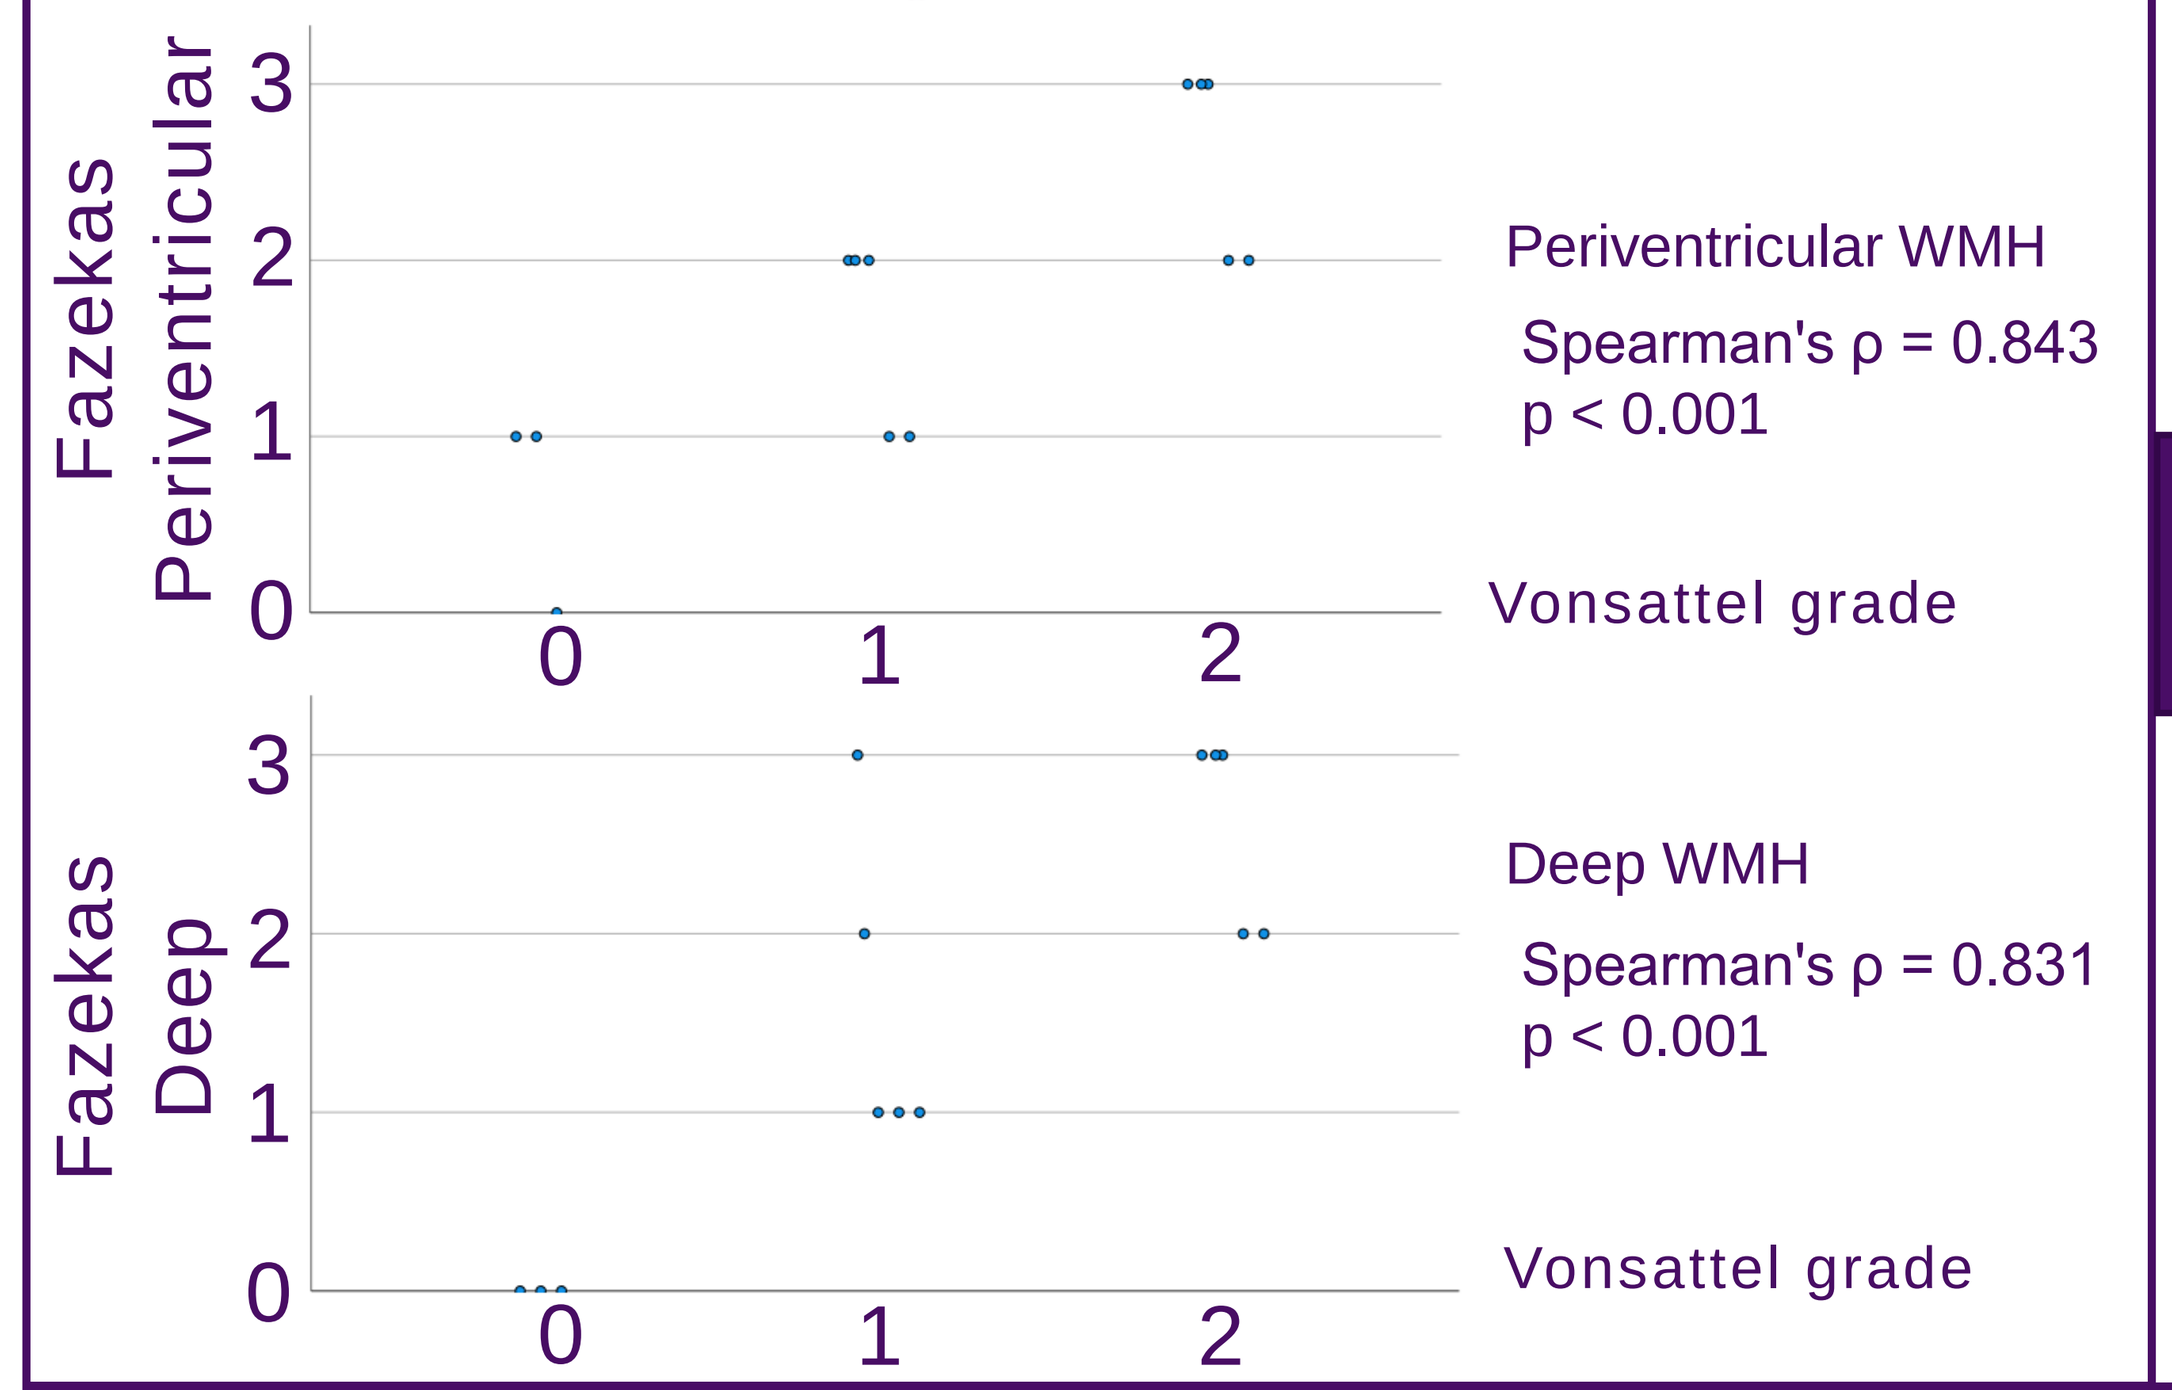
Fazekas Periventricular
3
2
1
0
0
1
2
Periventricular WMH
Spearman's ρ = 0.843
p < 0.001
Vonsattel grade
Fazekas Deep
Deep WMH
Spearman's ρ = 0.831
p < 0.001
Vonsattel grade

CONCLUSIONS

Pathological CAA severity was associated with WMH burden, but not with composite MRI SVD scores. Detailed Fazekas grading showed stronger pathological correlations than dichotomized WMH assessment.

Among patients with limited hemorrhagic MRI findings, detailed WMH assessment may provide complementary information beyond composite MRI SVD scores for estimating pathological CAA severity.

This approach may help refine MRI-based ARIA risk stratification in patients considered for anti-amyloid-β antibody therapy.

REFERENCES

1. Vonsattel JPG, et al. Ann Neurol 1991
2. Cummings J, et al. J Prev Alzheimers Dis 2023
3. Charidimou A, et al. Lancet Neurol 2022
4. Staals J, et al. Neurology 2014
5. Klarenbeek P, et al. Stroke 2013
6. Charidimou A, et al. JAMA Neurol 2016
7. Matsuda K, et al. BMJ Open 2021
8. Fazekas F, et al. AJR Am J Roentgenol 1987

Funding

This study was supported by JSPS KAKENHI (Grant No. JP24K02366) and conducted as part of a collaborative research agreement with Eli Lilly and Company.

CONTACT

Akihiro Shindo, MD, PhD

Department of Neurology,

Mie University Graduate School of Medicine, Tsu, Mie Japan

a-shindo@med.mie-u.ac.jp