Refer-MCI: Enriching referrals of patients to memory clinics for mild cognitive impairment (MCI)

Emrah Düzel[1], Jochen René Thyrian[2], Vanessa Kurth[3], Nadine Diersch[4], Anne Boehlen[5], Hannes Vossfeldt[4], Agnes Flöel[2], Anja Schneider[5], Wenzel Glanz[1], Kathrin Reetz[6], Jörg B. Schulz[6], Christine von Arnim[7], Lucrezia Hausner[8], Johannes Levin[9], Jens Wiltfang[10], David Berron[1], Frank Jessen[11], Annika Spottke[5], Stefan Teipel[12]

[1] German Center for Neurodegenerative Diseases (DZNE), Magdeburg, Germany · [2] DZNE, Greifswald, Germany · [3] Lilly Germany GmbH, Bad Homburg, Germany · [4] neotiv GmbH, Magdeburg, Germany · [5] DZNE, Bonn, Germany · [6] Department of Neurology, RWTH Aachen University, Germany · [7] Department of Geriatrics, University Medicine Göttingen, Germany · [8] Department of Gerontopsychiatry, Central Institute of Mental Health, Mannheim, Germany · [9] DZNE, Munich, Germany · [10] DZNE, Göttingen, Germany · [11] DZNE, Cologne, Germany · [12] DZNE, Rostock, Germany

Background

Timely diagnosis of mild cognitive impairment (MCI) due to Alzheimer's disease (AD) is crucial for early interventions, but its implementation in routine care is often challenging due to the complexity and time burden of required cognitive assessments. Currently, <10 % of people with MCI are identified in primary care in Germany (Bohlken & Kostev, 2019). Digital, remote, unsupervised (completely independent at home) self-testing of cognition can potentially address this health care challenge and may improve referral efficiency to memory clinics.

Goal

Evaluate whether the use of the CE-certified digital self-assessment app **neotivCare** can improve the accuracy and timeliness of referrals of people with suspected **mild cognitive impairment (MCI)** from outpatient specialist care to memory clinics in Germany. neotivCare's feasibility for the unsupervised, remote MCI detection in routine care has been previously investigated in the German healthcare system (Düzel et al., 2024; Blotenberg et al., 2025), providing first evidence for its feasibility and added value.

(1)

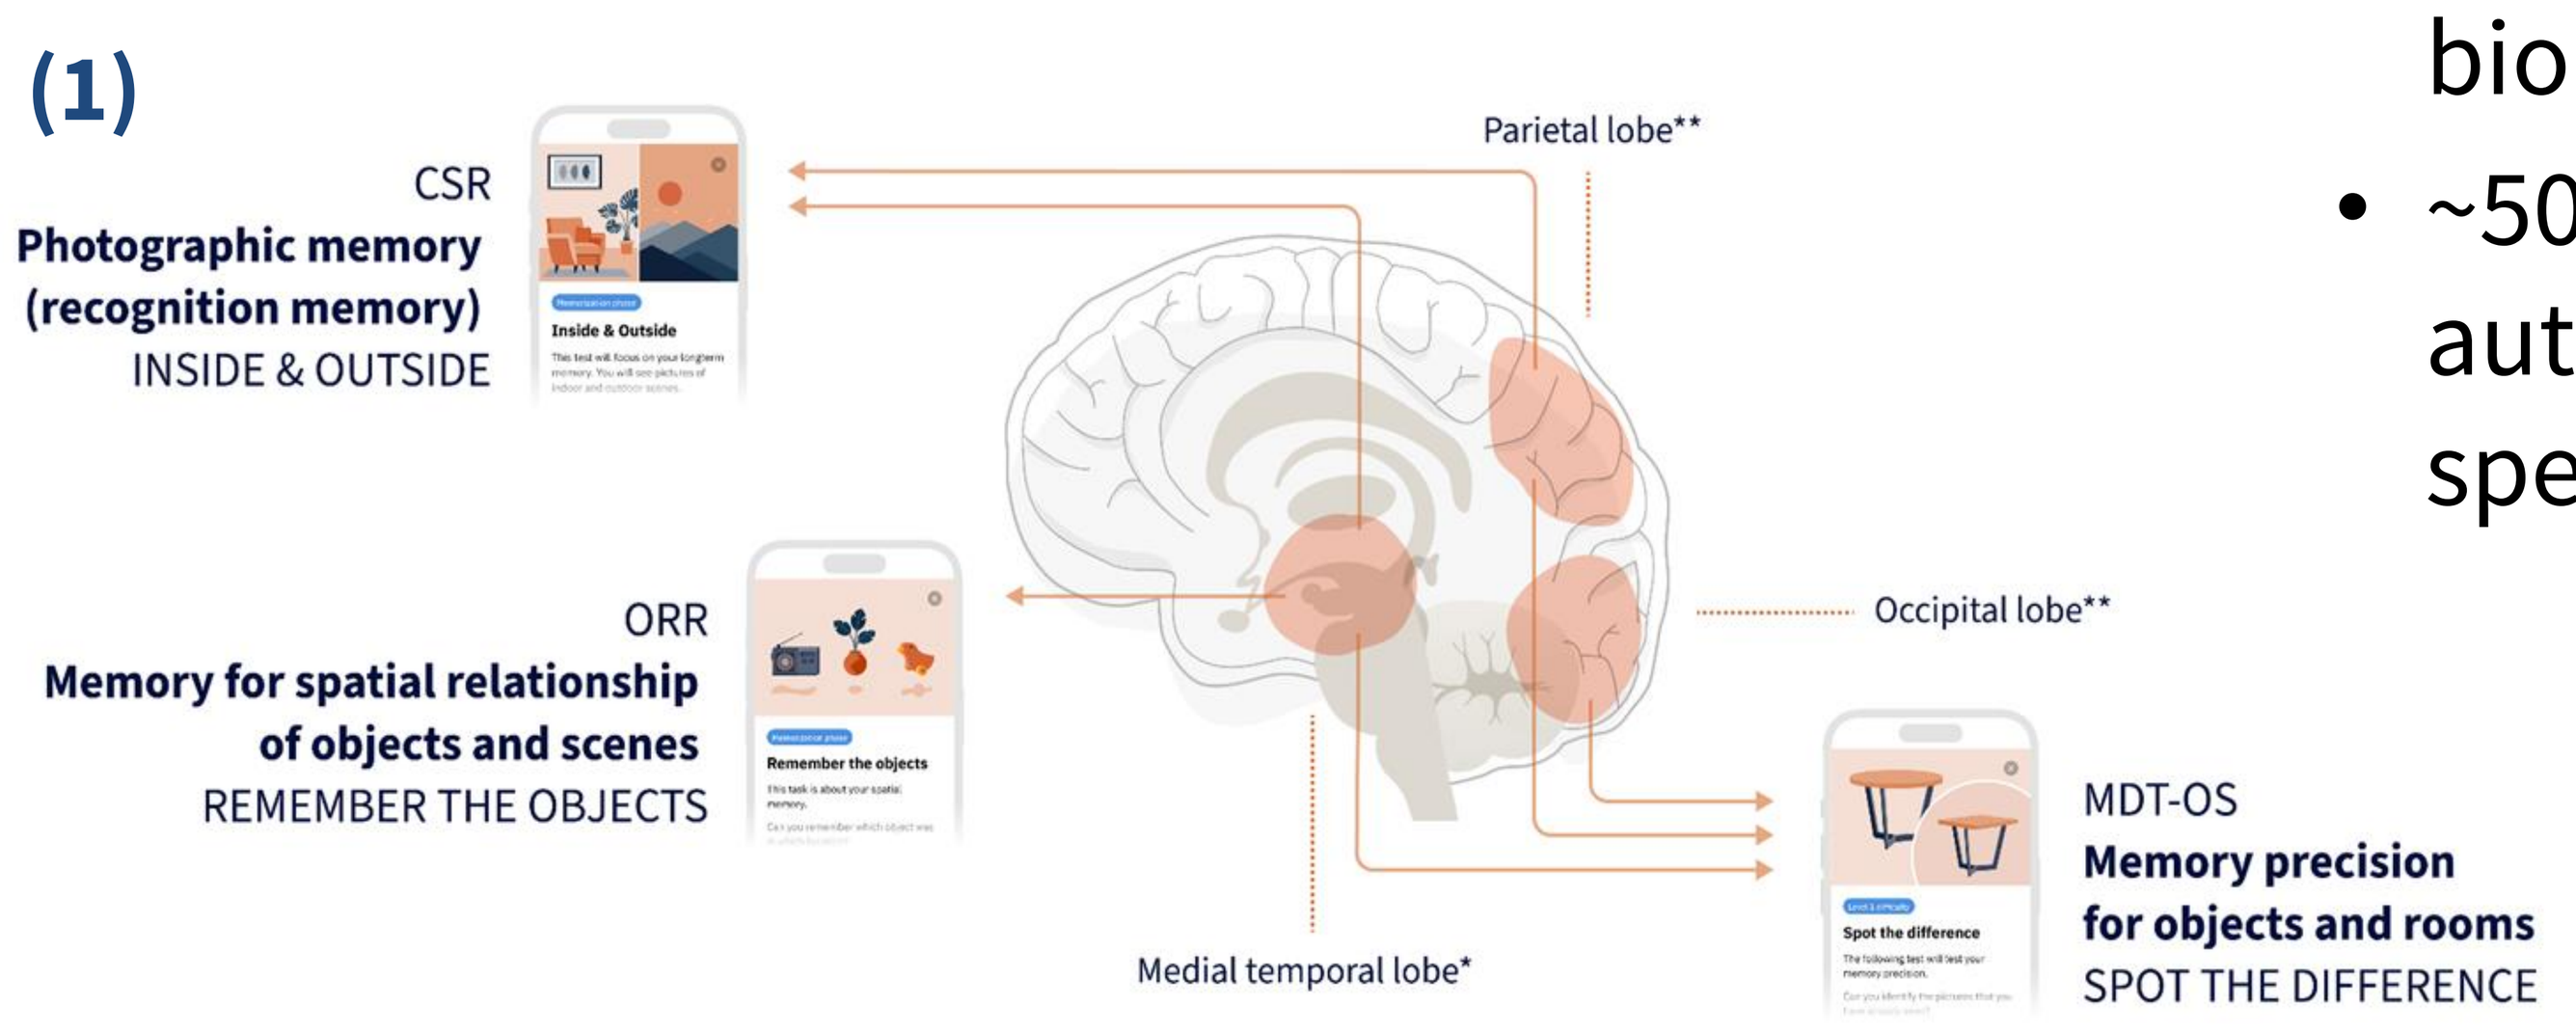
CSR
Photographic memory (recognition memory)
INSIDE & OUTSIDE
Inside & Outside
Parietal lobe**
ORR
Memory for spatial relationship of objects and scenes
REMEMBER THE OBJECTS
Remember the objects
Occipital lobe**
MDT-OS
Memory precision for objects and rooms
SPOT THE DIFFERENCE
Spot the difference
Medial temporal lobe*

* Areas of the medial temporal lobe: hippocampus and entorhinal, perirhinal and parahippocampal cortex
** Areas of spatial cognition (parietal lobe) and visual areas (occipital lobe)

Study Design

- Single-arm, multicentric healthcare study
- ~25 outpatient **neurological/psychiatric practices**
- 10 university **memory clinics** (DZNE- and DNG-associated)
- 500 participants aged 60–80 with self- or informant-reported memory complaints, referral for reference assessment (S3 guideline-based, Fig. 4)
- Planned duration: **21 months, FPI:** September 2025

Intervention

- Participants remote-tested at home using **neotivCare** weekly over 12 weeks.
- Referral decisions documented **before and after** app results.
- **All** participants referred to memory clinics for standardized assessment (CERAD, MMSE) and blood biomarker P-tau217 (Fig. 5).
- ~50 % subsample with automated phone-based speech biomarker assessment

(2)

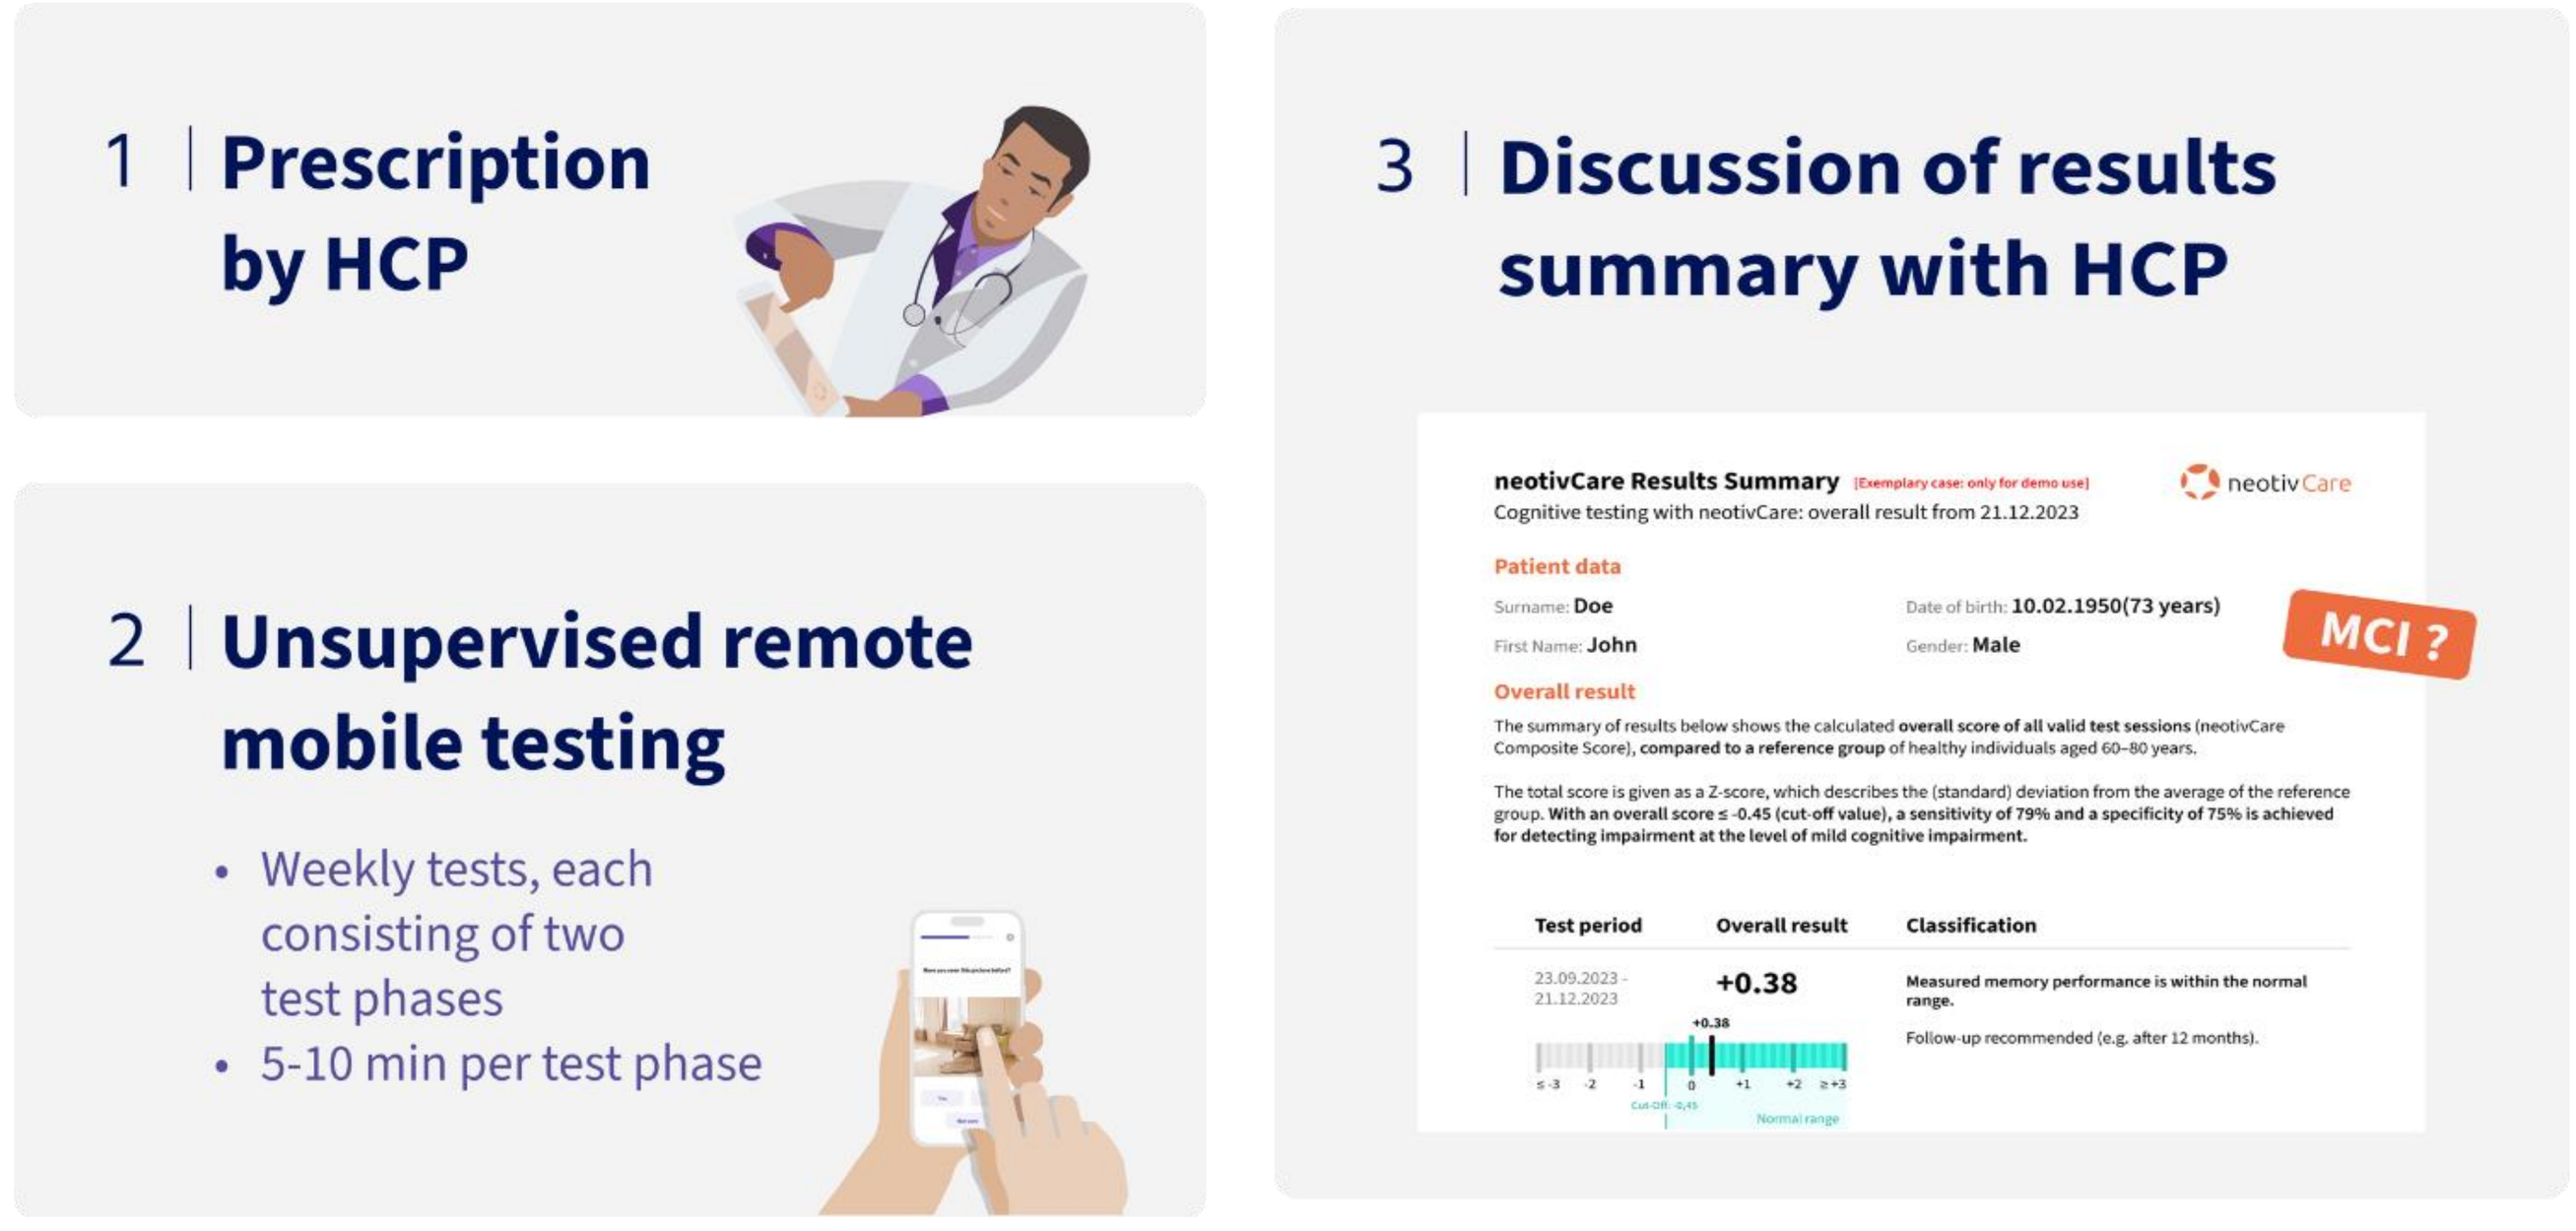
1 | Prescription by HCP
2 | Unsupervised remote mobile testing
• Weekly tests, each consisting of two test phases
• 5-10 min per test phase
3 | Discussion of results summary with HCP
neotivCare Results Summary
MCI ?
+0.38

(3)

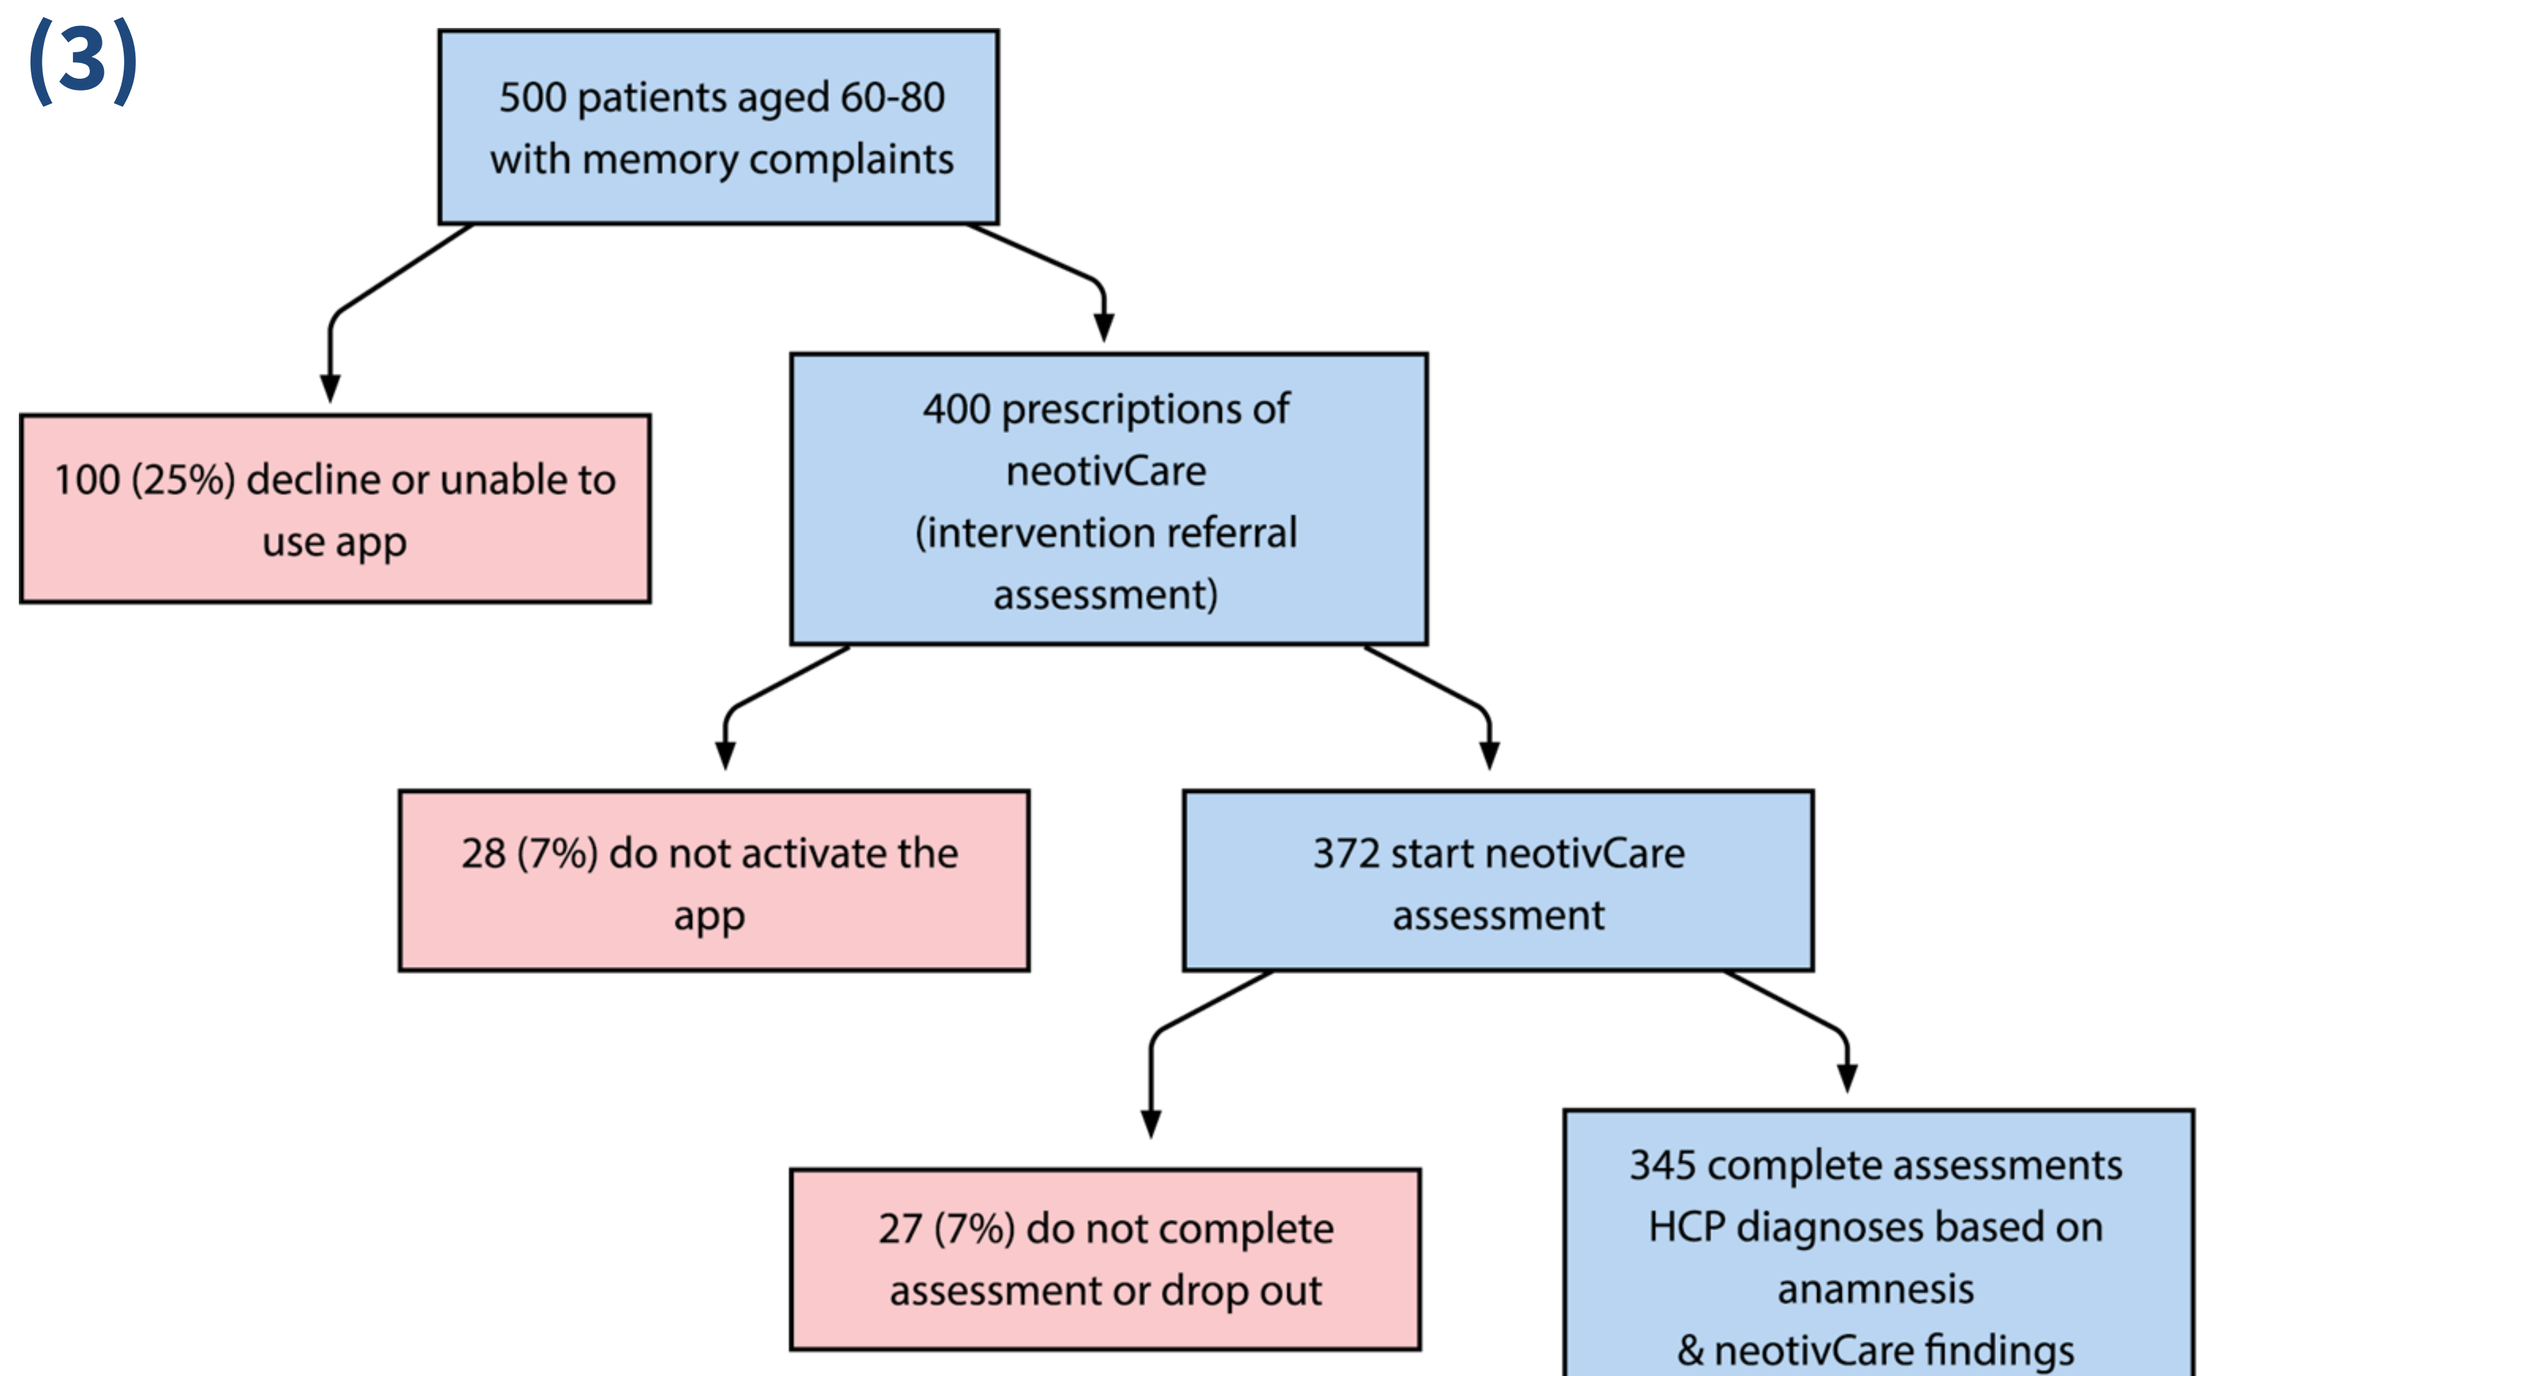
500 patients aged 60-80 with memory complaints
100 (25%) decline or unable to use app
400 prescriptions of neotivCare (intervention referral assessment)
28 (7%) do not activate the app
372 start neotivCare assessment
27 (7%) do not complete assessment or drop out
345 complete assessments HCP diagnoses based on anamnesis & neotivCare findings
15 drop out
330 patients (132 positive composite score) undergo reference assessment per S3 guidelines and pTau217
Final sample: N=120 with suspected MCI in intervention referral; N=320 for sensitivity/ specificity
Expected improvement of correct referrals

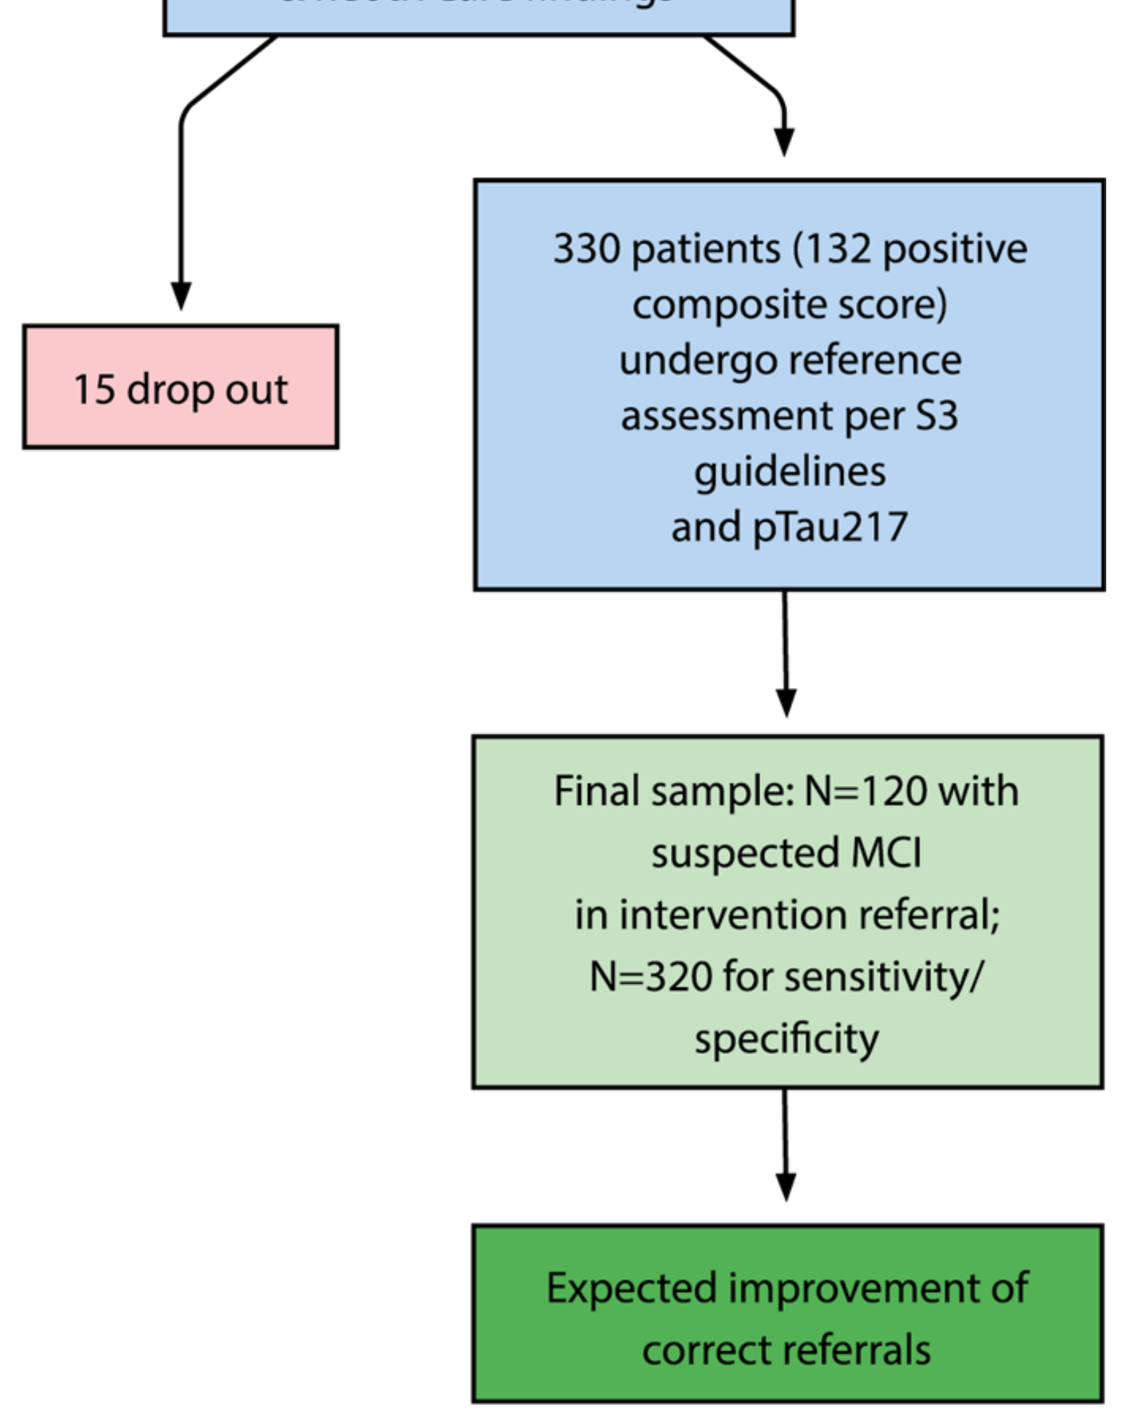

(4)

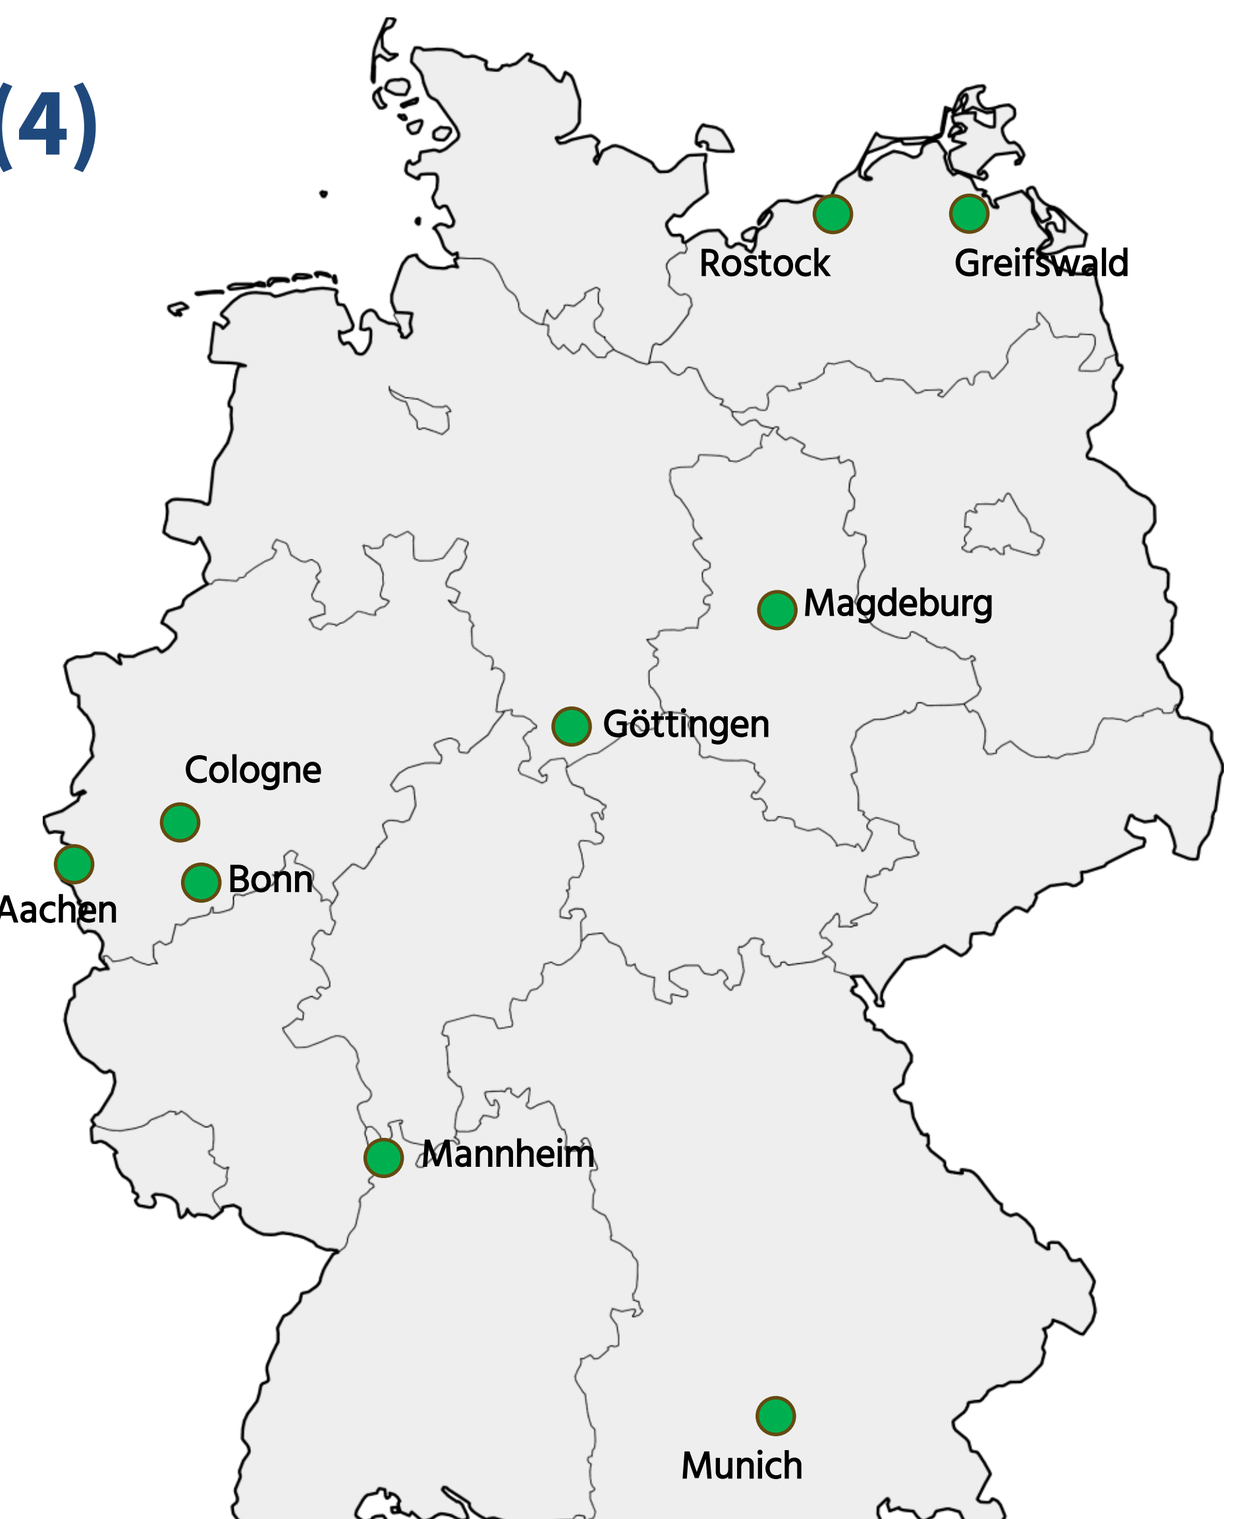
Rostock
Greifswald
Magdeburg
Göttingen
Cologne
Aachen
Bonn
Mannheim
Munich

(1) Anatomical specificity of the neotivCare tests
(2) neotivCare procedure
(3) Estimated patient flow in Refer-MCI

(4) Distribution of memory clinic study centers in Germany
(5) Recruitment status of (a) specialist offices and (b) patients (as of June 2026)
(6) Patient journey in Refer-MCI

Primary Endpoint

- Improvement in **correct referrals** (intervention vs. usual care), verified by **S3 guideline-based** memory clinic assessments.

Secondary Endpoints

- Sensitivity & specificity of referral decisions
- Proportion of MCI cases due to Alzheimer's disease (via **P-tau217**)
- Physician and patient feedback on usability, acceptance & barriers
- Feasibility of **speech biomarker analysis** via phone-based chatbot (ki:elements, ~50 % subsample)

Outcome Expectations

- 75 % participation rate (n = 400)
- improved referral accuracy using digital assessment
- Reduction in false-positive referrals
- Identification of barriers for implementation
- Feasibility of speech-based cognitive assessment in subsample

(5a)

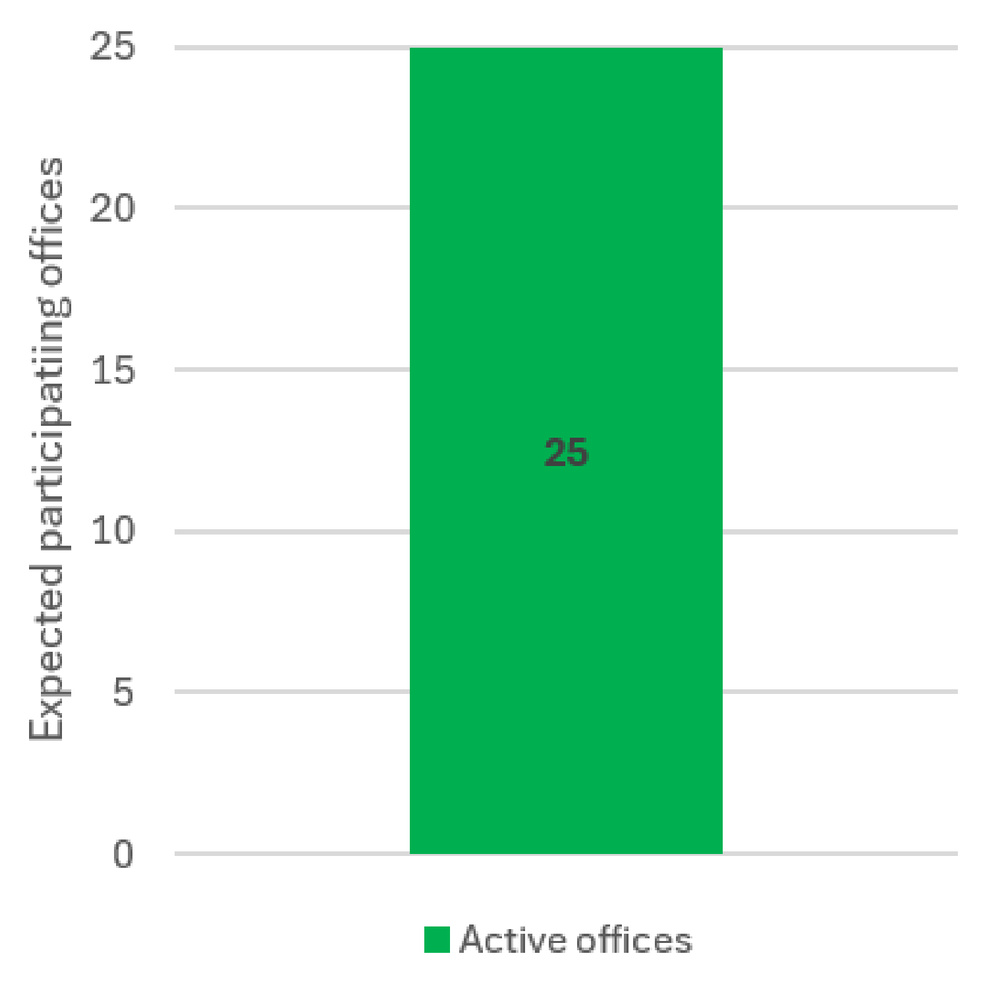
Expected participating offices
25
20
15
10
5
0
25
Active offices

(5b)

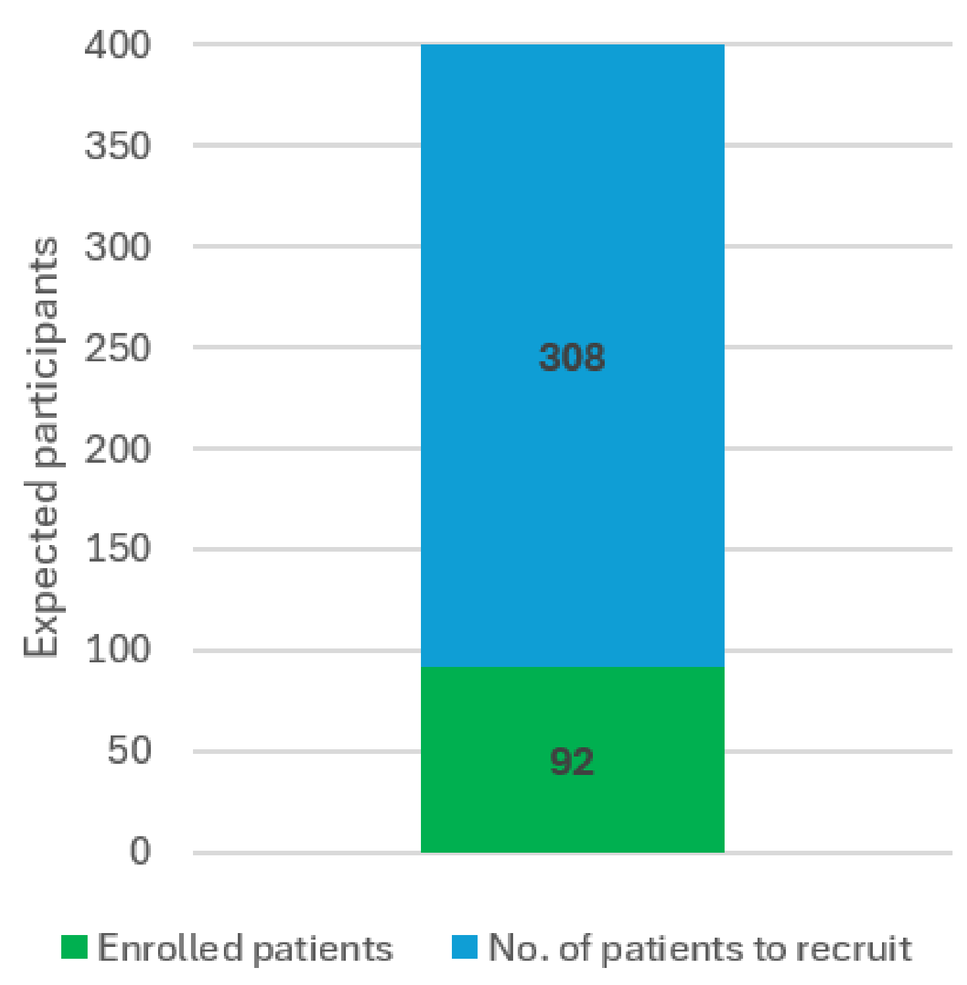
Expected participants
400
350
300
250
200
150
100
50
0
308
92
Enrolled patients
No. of patients to recruit

(6)

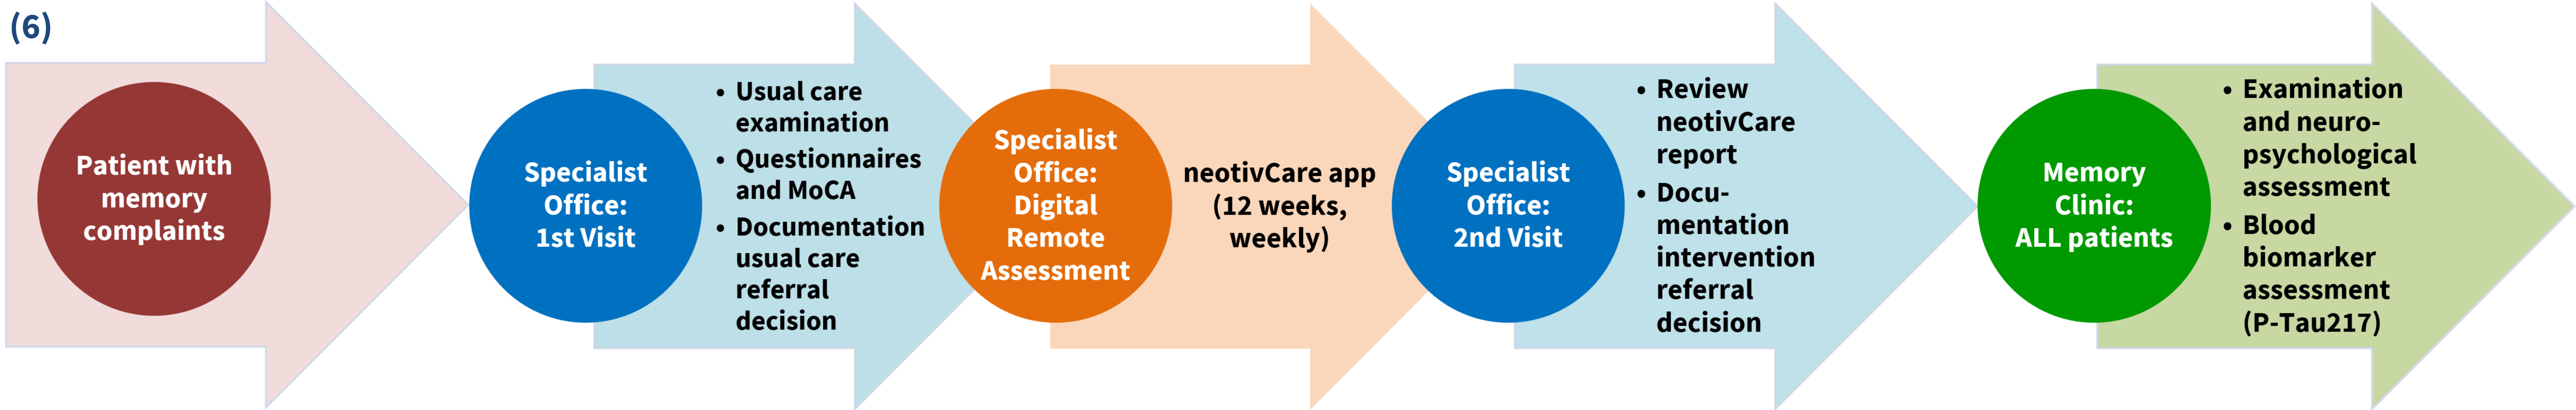
Patient with memory complaints
Specialist Office: 1st Visit
• Usual care examination
• Questionnaires and MoCA
• Documentation usual care referral decision
Specialist Office: Digital Remote Assessment
neotivCare app (12 weeks, weekly)
Specialist Office: 2nd Visit
• Review neotivCare report
• Docu-mentation intervention referral decision
Memory Clinic: ALL patients
• Examination and neuro-psychological assessment
• Blood biomarker assessment (P-Tau217)

In collaboration with

Lilly
A MEDICINE COMPANY

Preliminary Findings

The Refer-MCI project seeks to determine if remote digital cognitive assessments can aid in more timely detection and referral for diagnosis of MCI due to Alzheimer's disease. Early findings implementing app testing in specialist offices show that capacity challenges remain with opportunities to streamline onboarding training and improve medical assistant training and the need for even earlier referral from primary care offices.

1. Bohlken, J. & Kostev, K. (2019). Coded Prevalence of Mild Cognitive Impairment in General and Neuropsychiatrists Practices in Germany Between 2007 and 2017. Journal of Alzheimer's Disease, 1313–1318. https://doi.org/10.3233/jad-181180
2. Duzel, E., Schottler, M., Sommer, H., & Griebe, M. (2024). Protocol: Prospective evaluation of feasibility, added value and satisfaction of remote digital self-assessment for mild cognitive impairment in routine care with the neotivCare app. BMJ Open, 14(3), e081159. doi:10.1136/bmjopen-2023-081159
3. Blotenberg, I., Boekholt, M., Lieberknecht, N., Säring, P. & Thyrian, J. R. (2025). Acceptance of Unsupervised App-Based Cognitive Assessment in Outpatient Care: An Implementation Study. JMIR Formative Research, 9, e62706–e62706. https://doi.org/10.2196/62706