Tue-1061

Neurocognitive, Biomarker, and Health Outcomes in Those at Risk for Alzheimer's Disease Symptoms: ANCHOR-AD Study Design

Scan the QR code for a list of all Lilly content presented at the congress. Other company and product names are trademarks of their respective owners.

Sterling C Johnson[1], Nalin Payakachat[2], Michelle M Mielke[3], Kellen K Petersen[4], Alette Wessels[2], E Jolanda Muenzel[2], Dulanji K Kuruppu[2], Rose C Beck[2], J Scott Andrews[2], Yoshiki Niimi[5], Roy Yaari[2], Takeshi Iwatsubo[6], Suzanne E Schindler[4], Reisa Sperling[7]

[1]University of Wisconsin, Madison, USA, [2]Eli Lilly and Company, Indianapolis, USA, [3]Wake Forest University School of Medicine, Winston-Salem, USA, [4]Washington University School of Medicine, St. Louis, USA, [5]University of Tokyo Hospital, Tokyo, Japan, [6]University of Tokyo National Center of Neurology and Psychiatry, Tokyo, Japan, [7]Massachusetts General Hospital, Boston, USA

Sponsored by Eli Lilly and Company

OBJECTIVES

- ANCHOR-AD (NCT07142954) is designed to characterize clinical trajectories associated with baseline plasma P-tau217 in cognitively unimpaired individuals.
- This study may also inform which cognitively unimpaired individuals in the general population are likely to develop Alzheimer's disease (AD) pathology.

CONCLUSIONS

- ANCHOR-AD will enable further research to support the growing body of AD epidemiology, particularly the natural progression of the early stages of AD, and the management of individuals who are cognitively unimpaired with elevated plasma P-tau217 levels.
- The biobank from this study will enhance future biomarker studies for AD and other neurodegenerative diseases.

Alzheimer's Association International Conference 2026; London, United Kingdom; July 12-15, 2026

BACKGROUND

Biomarkers of AD:

- AD pathology can begin decades before individuals develop cognitive and functional symptoms.
- Biomarker evidence of AD pathology can be detected in cognitively unimpaired individuals.
- Phosphorylated tau at position 217 (P-tau217) is a blood biomarker that reliably detects AD pathology and is associated with faster cognitive decline and increased progression risk in cognitively unimpaired individuals.

Real-World Data:

- Prior studies have generated foundational knowledge on the natural history of cognitively unimpaired (preclinical) AD and the prognostic value of blood biomarkers by demonstrating significant associations between biomarkers and cognitive decline.
- A need exists to generate large-scale, longitudinal, real-world data integrating plasma P-tau217 with cognitive assessments across diverse populations.
- ANCHOR-AD is designed to address this gap by evaluating the prognostic utility of plasma P-tau217 and generating population-level data on cognitive trajectories and blood biomarkers in cognitively unimpaired individuals.

STUDY OBJECTIVES & ENDPOINTS

Primary:

- Determine rates of cognitive worsening within elevated and not elevated plasma P-tau217 cohorts (measured as change from baseline in the cognitive composite or any individual components).

Secondary:

- Compare rates of cognitive worsening between elevated and not elevated plasma P-tau217 cohorts (measured as change from baseline in the cognitive composite or any individual components).
- Characterize time to and rate of cognitive worsening within each cohort on different measures (ISLT, iDSSTm, CPAL, CFI, Xpresso).
- Develop a predictive model of progression to cognitive worsening based on participant characteristics.
- Establish the frequency of elevated plasma P-tau217 in cognitively unimpaired adults in the US and Japan.

Exploratory:

- Measure progression rate from not elevated to elevated plasma P-tau217 and develop a predictive model of biomarker change based on participant characteristics.

Community-Based Recruitment

- **US:** Single virtual site led by Care Access; recruitment via Care Access's Future of Medicine Program (registry of potential participants) and Referral Program (referral pathway into clinical studies)
- **Japan:** Three clinical sites; recruitment through the University of Tokyo Hospital and Japan-Trial Ready Cohort – Japan National Registry

Enrollment & Cohort Stratification

- N ≈ 2,200 individuals in the US (equal split between cohorts)
- N ≈ 1,200 individuals in Japan (equal split between cohorts)

Elevated P-tau217:

Baseline P-tau217 ≥ cutoff determined using the Roche Elecsys assay

Stratified by age (55-64, 65-69, and 70-75 yrs) and geographic location

Not Elevated P-tau217:

Baseline P-tau217 < cutoff determined using the Roche Elecsys assay

Stratified by age (55-64, 65-69, and 70-75 yrs) and geographic location

Classified into intermediate and low sub-cohorts

Building Blocks of ANCHOR-AD

Scale
~3,400 participants in US + Japan

Duration
7-year follow-up

Real-World Data Linkage
Optional EHR + health insurance claims broaden data (US)

Culturally Adapted Tests
Cognitive battery (ISLT, CPAL, CFI, iDSSTm, XpressO); validated for US + Japan

Decentralized Design
Virtual assessments and local blood draws

Diversity
Community-based enrollment (US)
Three clinical sites (Japan)

Annual Plasma P-tau217
Roche Elecsys assay + additional biosample collection and biobank generation

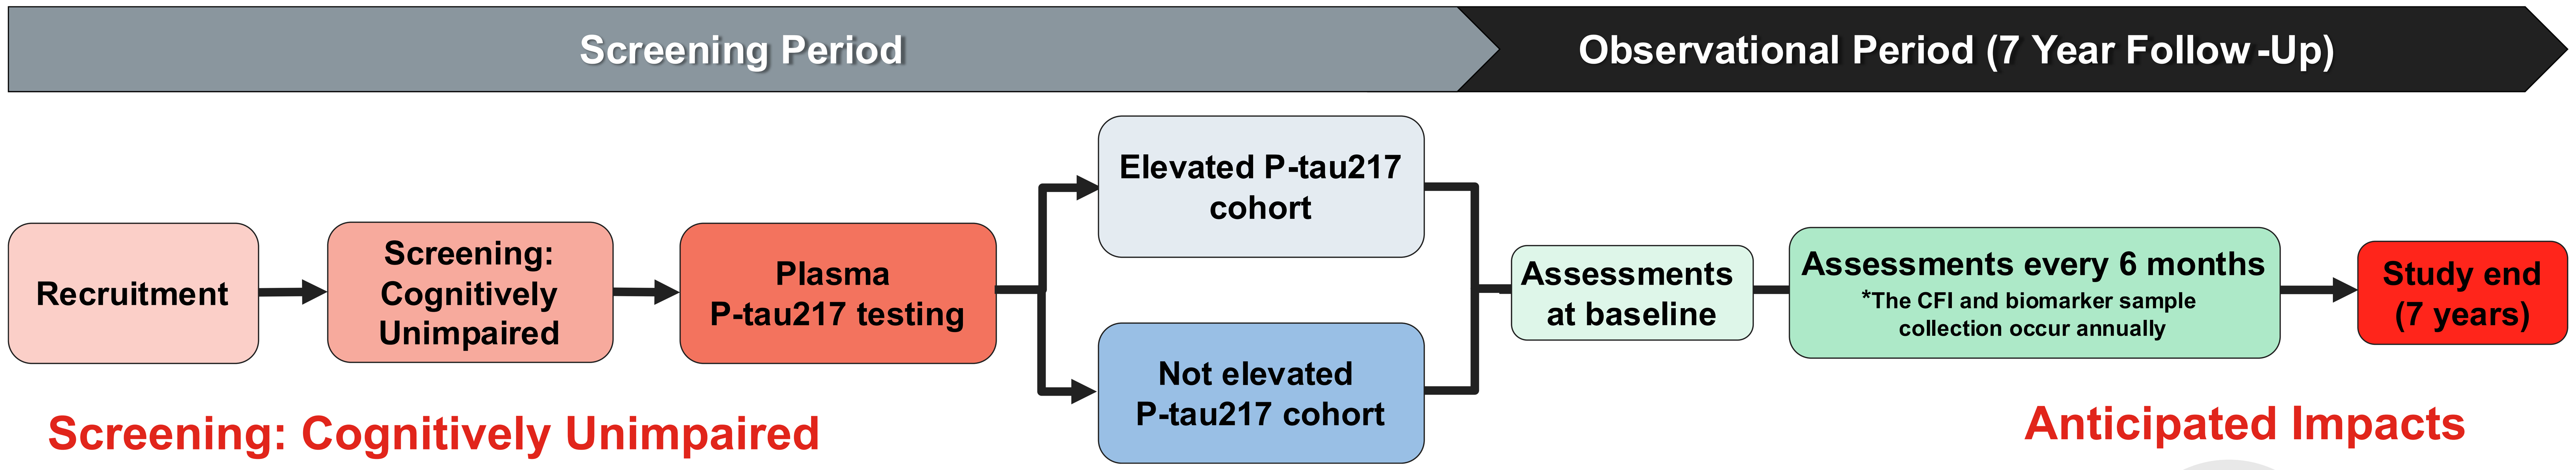

Screening Period
Observational Period (7 Year Follow-Up)
Recruitment
Screening: Cognitively Unimpaired
Plasma P-tau217 testing
Elevated P-tau217 cohort
Not elevated P-tau217 cohort
Assessments at baseline
Assessments every 6 months
*The CFI and biomarker sample collection occur annually
Study end (7 years)

Screening: Cognitively Unimpaired

- N ≈ 15,000 individuals in the US
- N ≈ 9,000 individuals in Japan

Key Inclusion Criteria

- 55 – 75 years of age
- Self-report unimpaired cognition
- Adequate literacy, vision, and hearing for assessments at screening
- Access to internet or cellular service and a device compatible with assessments

Key Exclusion Criteria

- Seen a doctor about memory concerns
- History of cognitive impairment
- Received or receiving treatment for AD (e.g., investigational or amyloid-targeting therapy)
- Enrolled or participated in any AD-related study

Schedule of Assessments

Assessments and Activities	Baseline	Every 6 months	Every 12 months
Plasma P-tau217 Assay	X (screening visit)		X
Apolipoprotein E Genotype	X		
Biomarker Sample Collection	X		X
Self-Reported Comorbidities and Medication Use	X	X	
Cognitive Function Index	X		X
Continuous Paired Associate Learning Test	X	X	
Hospital Anxiety and Depression Scale	X	X	
International Daily Symbol Substitution Test-Medicines	X	X	
International Shopping List Test	X	X	
Montreal Cognitive Assessment XpressO	X	X	
Quick Dementia Rating System	X	X	
Resource Use Inventory-Brief	X	X	

Anticipated Impacts

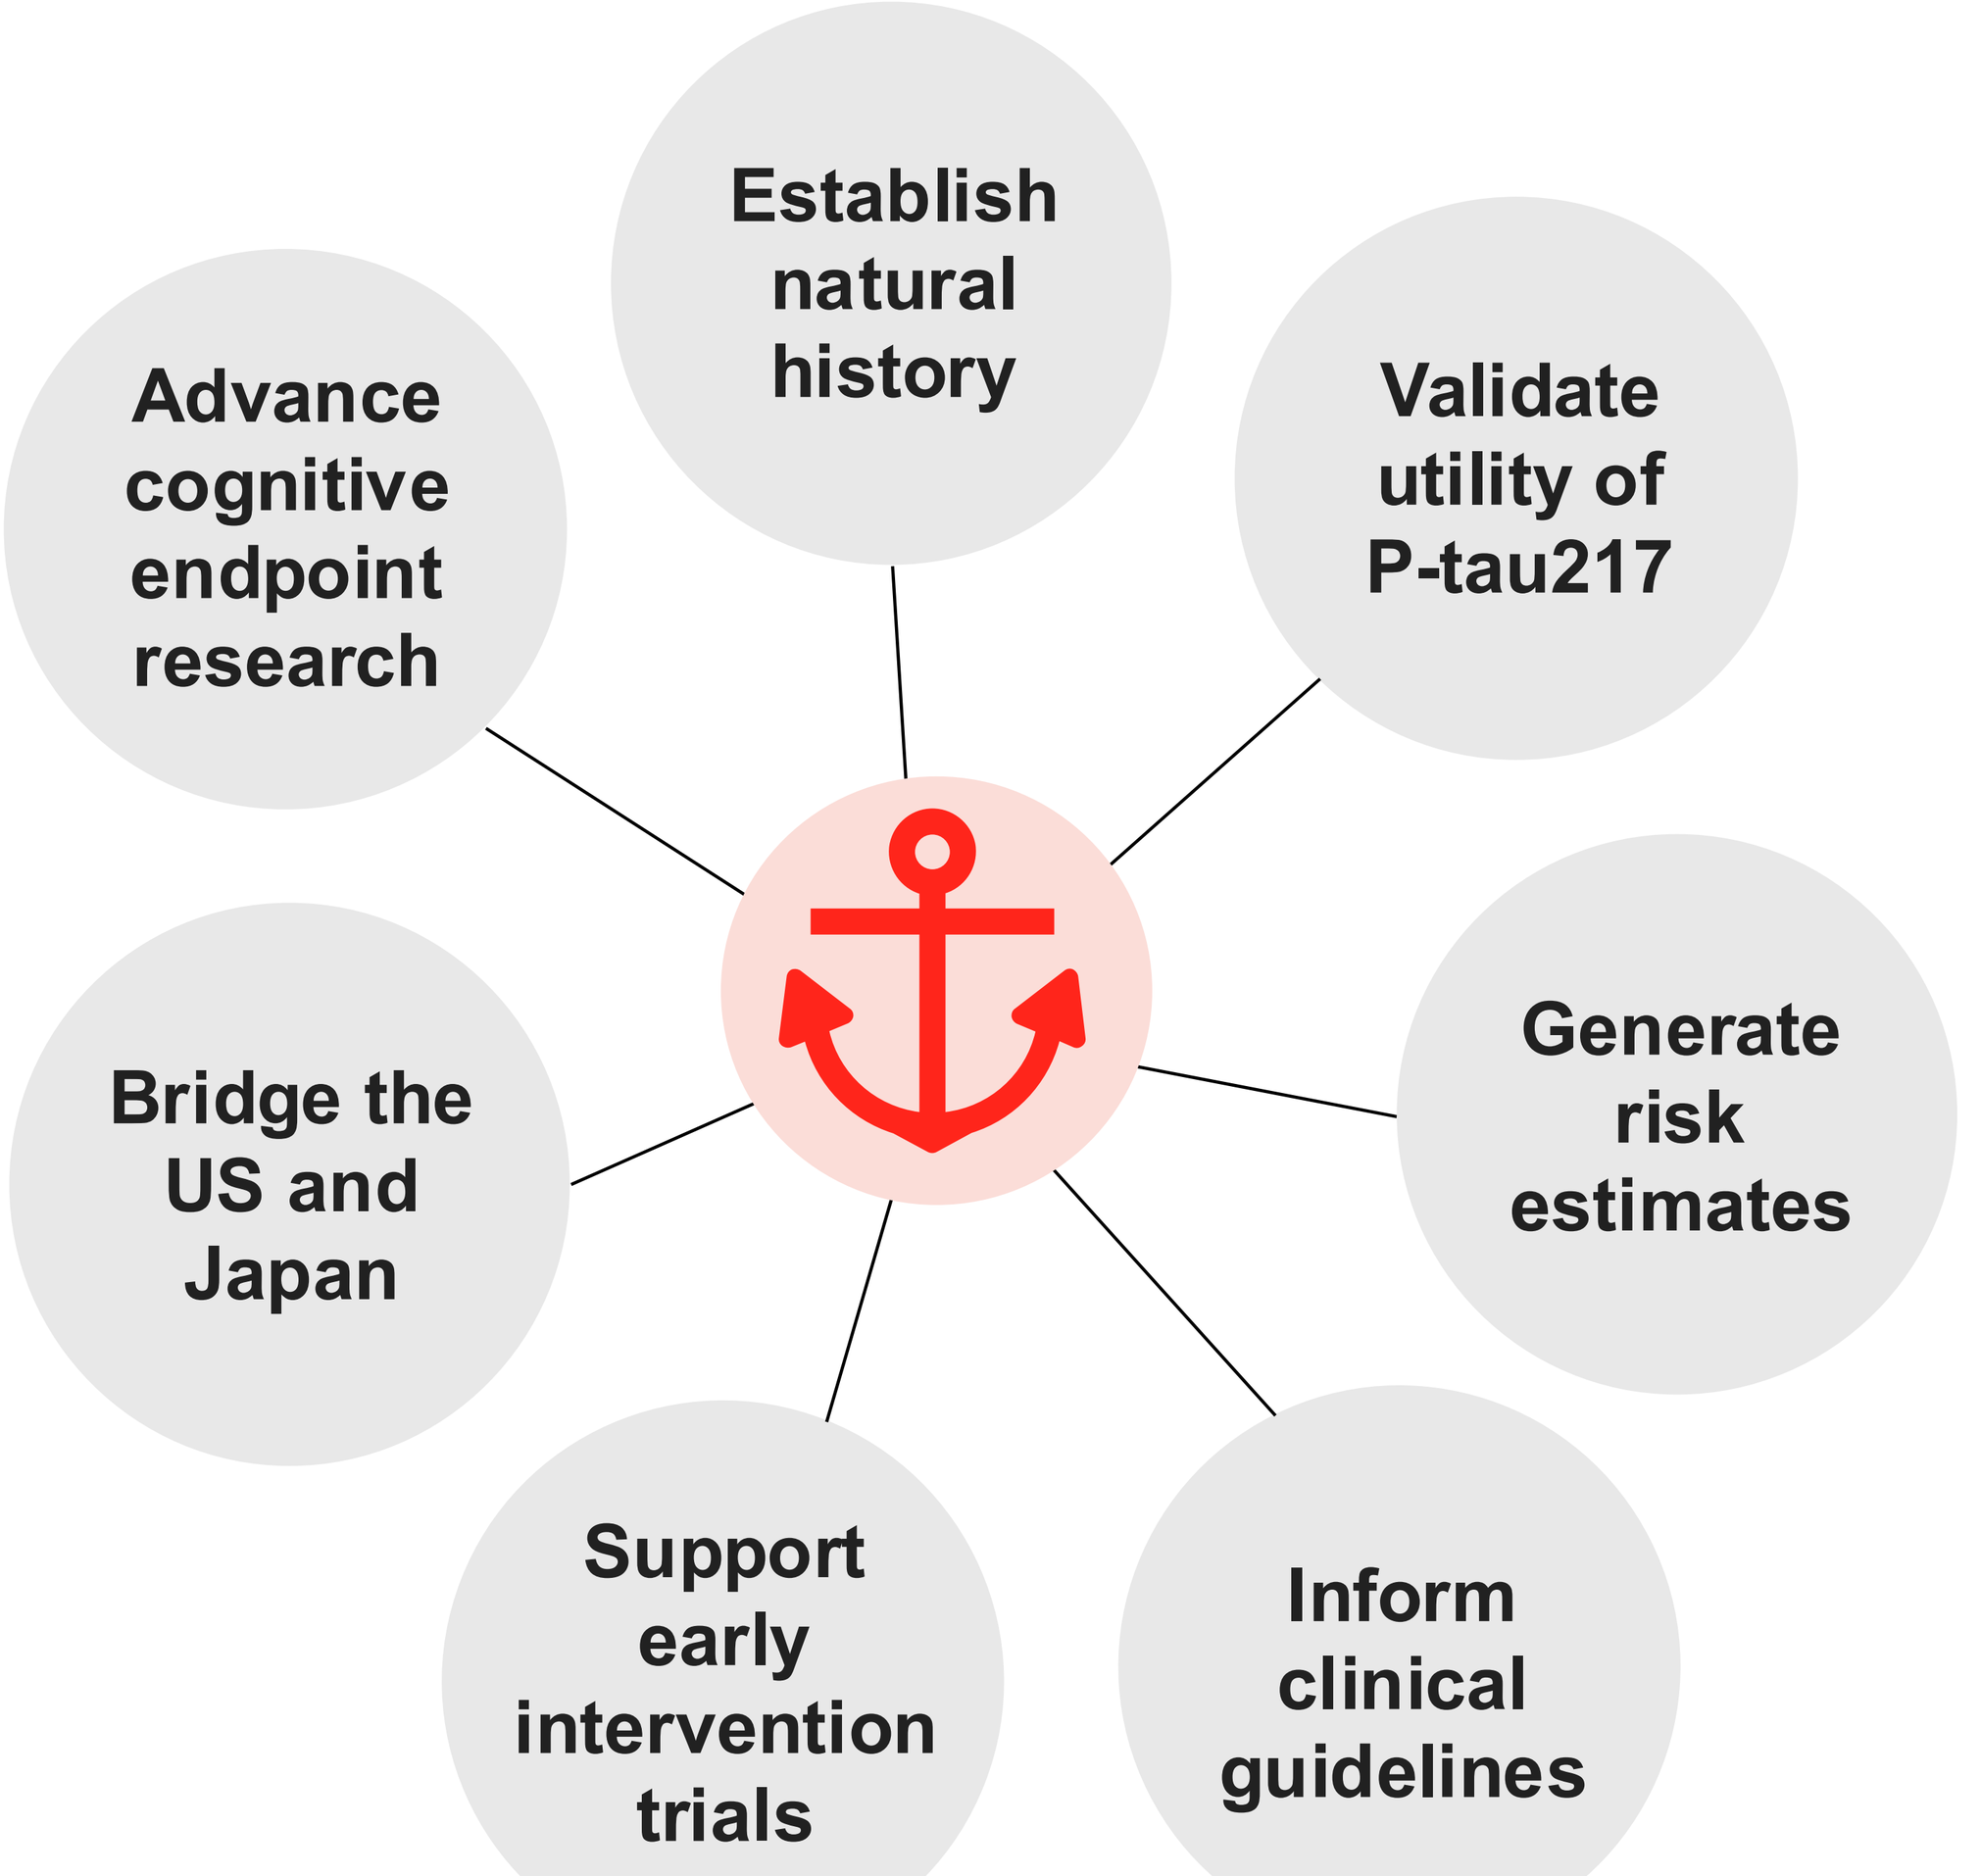

Establish natural history
Validate utility of P-tau217
Generate risk estimates
Inform clinical guidelines
Support early intervention trials
Bridge the US and Japan
Advance cognitive endpoint research

References

1. Jack, C.R., Jr., et al., Revised criteria for diagnosis and staging of Alzheimer's disease: Alzheimer's Association Workgroup. Alzheimers Dement, 2024. 20(8): p. 5143-5169.
2. Ashton, N.J., et al., Diagnostic Accuracy of a Plasma Phosphorylated Tau 217 Immunoassay for Alzheimer Disease Pathology. JAMA Neurology, 2024. 81(3): p. 255.
3. Mattsson-Carlgren, N., et al., Prediction of Longitudinal Cognitive Decline in Preclinical Alzheimer Disease Using Plasma Biomarkers. JAMA Neurol, 2023. 80(4): p. 360-369.
4. Jessen, F., et al., Design and first baseline data of the DZNE multicenter observational study on predementia Alzheimer's disease (DELCODE). Alzheimers Res Ther, 2018. 10(1): p. 15.
5. Johnson, S.C., et al., The Wisconsin Registry for Alzheimer's Prevention: A review of findings and current directions. Alzheimers Dement (Amst), 2018. 10: p. 130-142.

Disclosures: SCJ served as a consultant to Enigma Biomedical, Eli Lilly and Company, and AlzPath. NP, AW, EJM, DK, RCB, JSA, and RY are employees and minor shareholders of Eli Lilly and Company. MMM has served as an advisor and/or has consultant for Althira, Biogen, Cognito Therapeutics, Eisai, LabCorp, Eli Lilly and Company, Merck, Neurogen Biomarking, Novo Nordisk, Roche, and Siemens Healthineers, and received speaking honorariums from Novo Nordisk, PeerView Institute, and Roche. KKP has served as a consultant for Eli Lilly and Company. YN received consultancy/speaker fees from Eli Lilly and Company. TI received consultancy/speaker fees from Eli Lilly and Company. SES has served as an advisor or consultant, or received speaker fees from Eisai, Novo Nordisk, Medscape, PeerView, Academy for Continued Healthcare Learning, Acumen, Biogen, Cognito Therapeutics, Danaher, Johnson and Johnson Innovative Medicine, Sanofi, and Eli Lilly and Company. RS has received funding to trial sites from Eli Lilly and Company and Eisai; consultant fees from AbbVie, AC Immune, Acumen, Alector, Apellis, Biohaven, Bristol Myers Squibb, Genentech, Ionis, Janssen, Nervgen, Oligomerix, Prothena, Roche, Vigis Neuroscience, and Vaxxinity; and travel reimbursement from the Alzheimer's Association, Clinical Trials in Alzheimer's Disease, and Janssen.

Acknowledgments: The authors would like to thank Lindsey Kennedy, PhD, of Eli Lilly and Company, for their writing and editorial contributions.

Abbreviations: AD = Alzheimer's disease; CFI = Cognitive Function Index; CPAL = Continuous Paired Associate Learning Test; EHR = Electronic Health Records; iDSSTm = International Daily Symbol Substitution Test-Medicines; ISLT = International Shopping List Test; P-tau217 = phosphorylated tau at threonine 217; US = United States; XpressO = Montreal Cognitive Assessment Xpresso

Copyright ©2026 Eli Lilly and Company. All rights reserved.