Native Methylation Sequencing for Detection and Monitoring of Alzheimer's Disease, Parkinson's, and Other Neurodegenerative Conditions

Author Block: Chad Pollard, PhD[1], Ryan Miller[1], Isaac Stirland[1], Mykle Keni[1], Jonathon Hill[2,1], Erin Saito[1], Tim Jenkins[2,1]

1Resonant, Pleasant Grove, UT, USA; 2Cell Biology and Physiology, Brigham Young University, Provo, UT, USA.

Background

Blood-based biomarkers are improving early detection and staging in neurodegenerative disease, but current assays primarily reflect aggregate pathology or generalized neuronal injury and provide limited insight into which neuronal populations are actively affected. Circulating cell-free DNA (cfDNA) offers a complementary biomarker substrate because it retains DNA methylation patterns linked to tissue and cellular origin, enabling cell-of-origin analysis from blood.

Here, we evaluated whether native nanopore methylation sequencing could support both brain methylation atlas generation and downstream cfDNA cell-of-origin classifier development in neurodegenerative disease. This framework was applied to Alzheimer's disease (AD), Parkinson's disease (PD), and amyotrophic lateral sclerosis (ALS) to test whether plasma cfDNA carries neuron-aligned methylation signals corresponding to selectively vulnerable neuronal populations

Methods

Whole-genome methylation reference profiles were generated from purified primary human neurons and glia and used to develop cell-type-informed cfDNA classifiers.

Sample Sources:
- Genomic DNA from primary human cortical, dopaminergic, and spinal motor neurons, and glial cells (astrocytes, microglia, Schwann cells)
- Plasma cfDNA from a filtered cohort of MCI/AD, PD, ALS, and healthy controls (n = 137)

Native Sequencing:
- Direct Whole Methylome Sequencing for gDNA (Wasatch BioLabs)
- Direct Methylation Sequencing for cfDNA (Wasatch BioLabs)
- Oxford Nanopore PromethION

Data Analysis:
- Differential methylation analysis and methylation-based cell-of-origin deconvolution
- In silico analytical validation, clinical plasma classification, and disease modeling

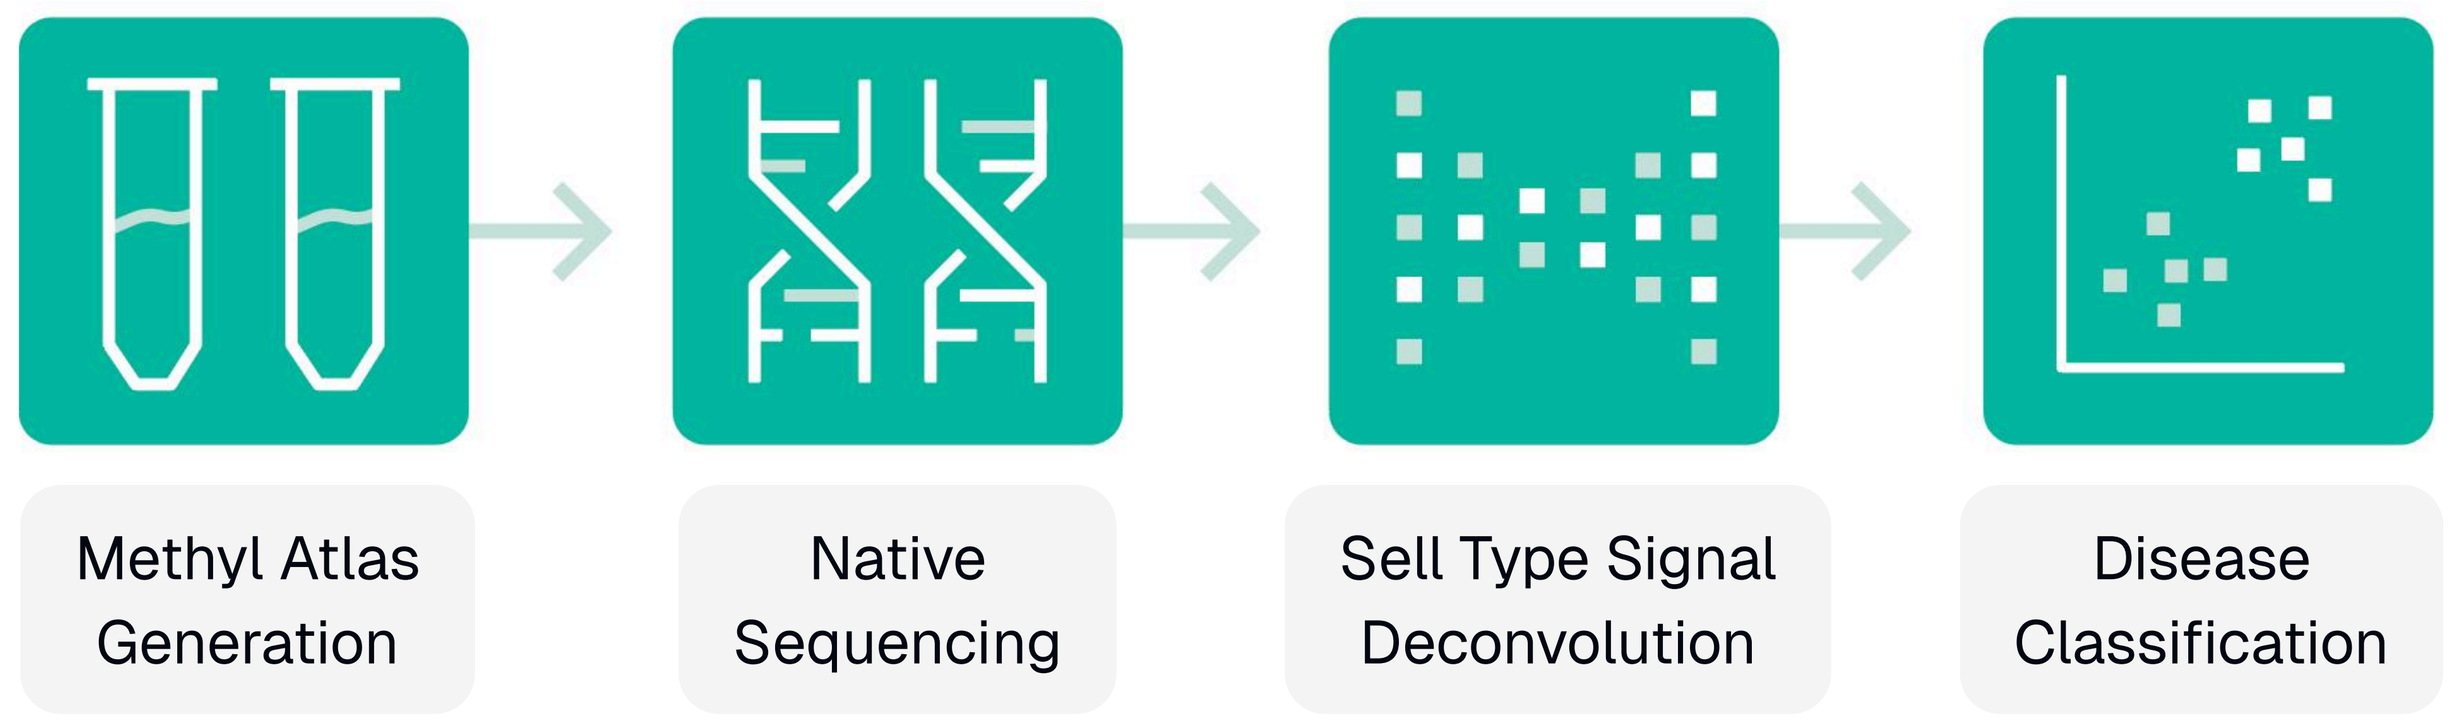
Methyl Atlas Generation
Native Sequencing
Sell Type Signal Deconvolution
Disease Classification

Figure 1. High-level study workflow. Workflow includes reference atlas generation, native sequencing, methylation-based cfDNA deconvolution, and disease classification.

Results

Predicting Cell Fractions at Cell-Associated DMRs

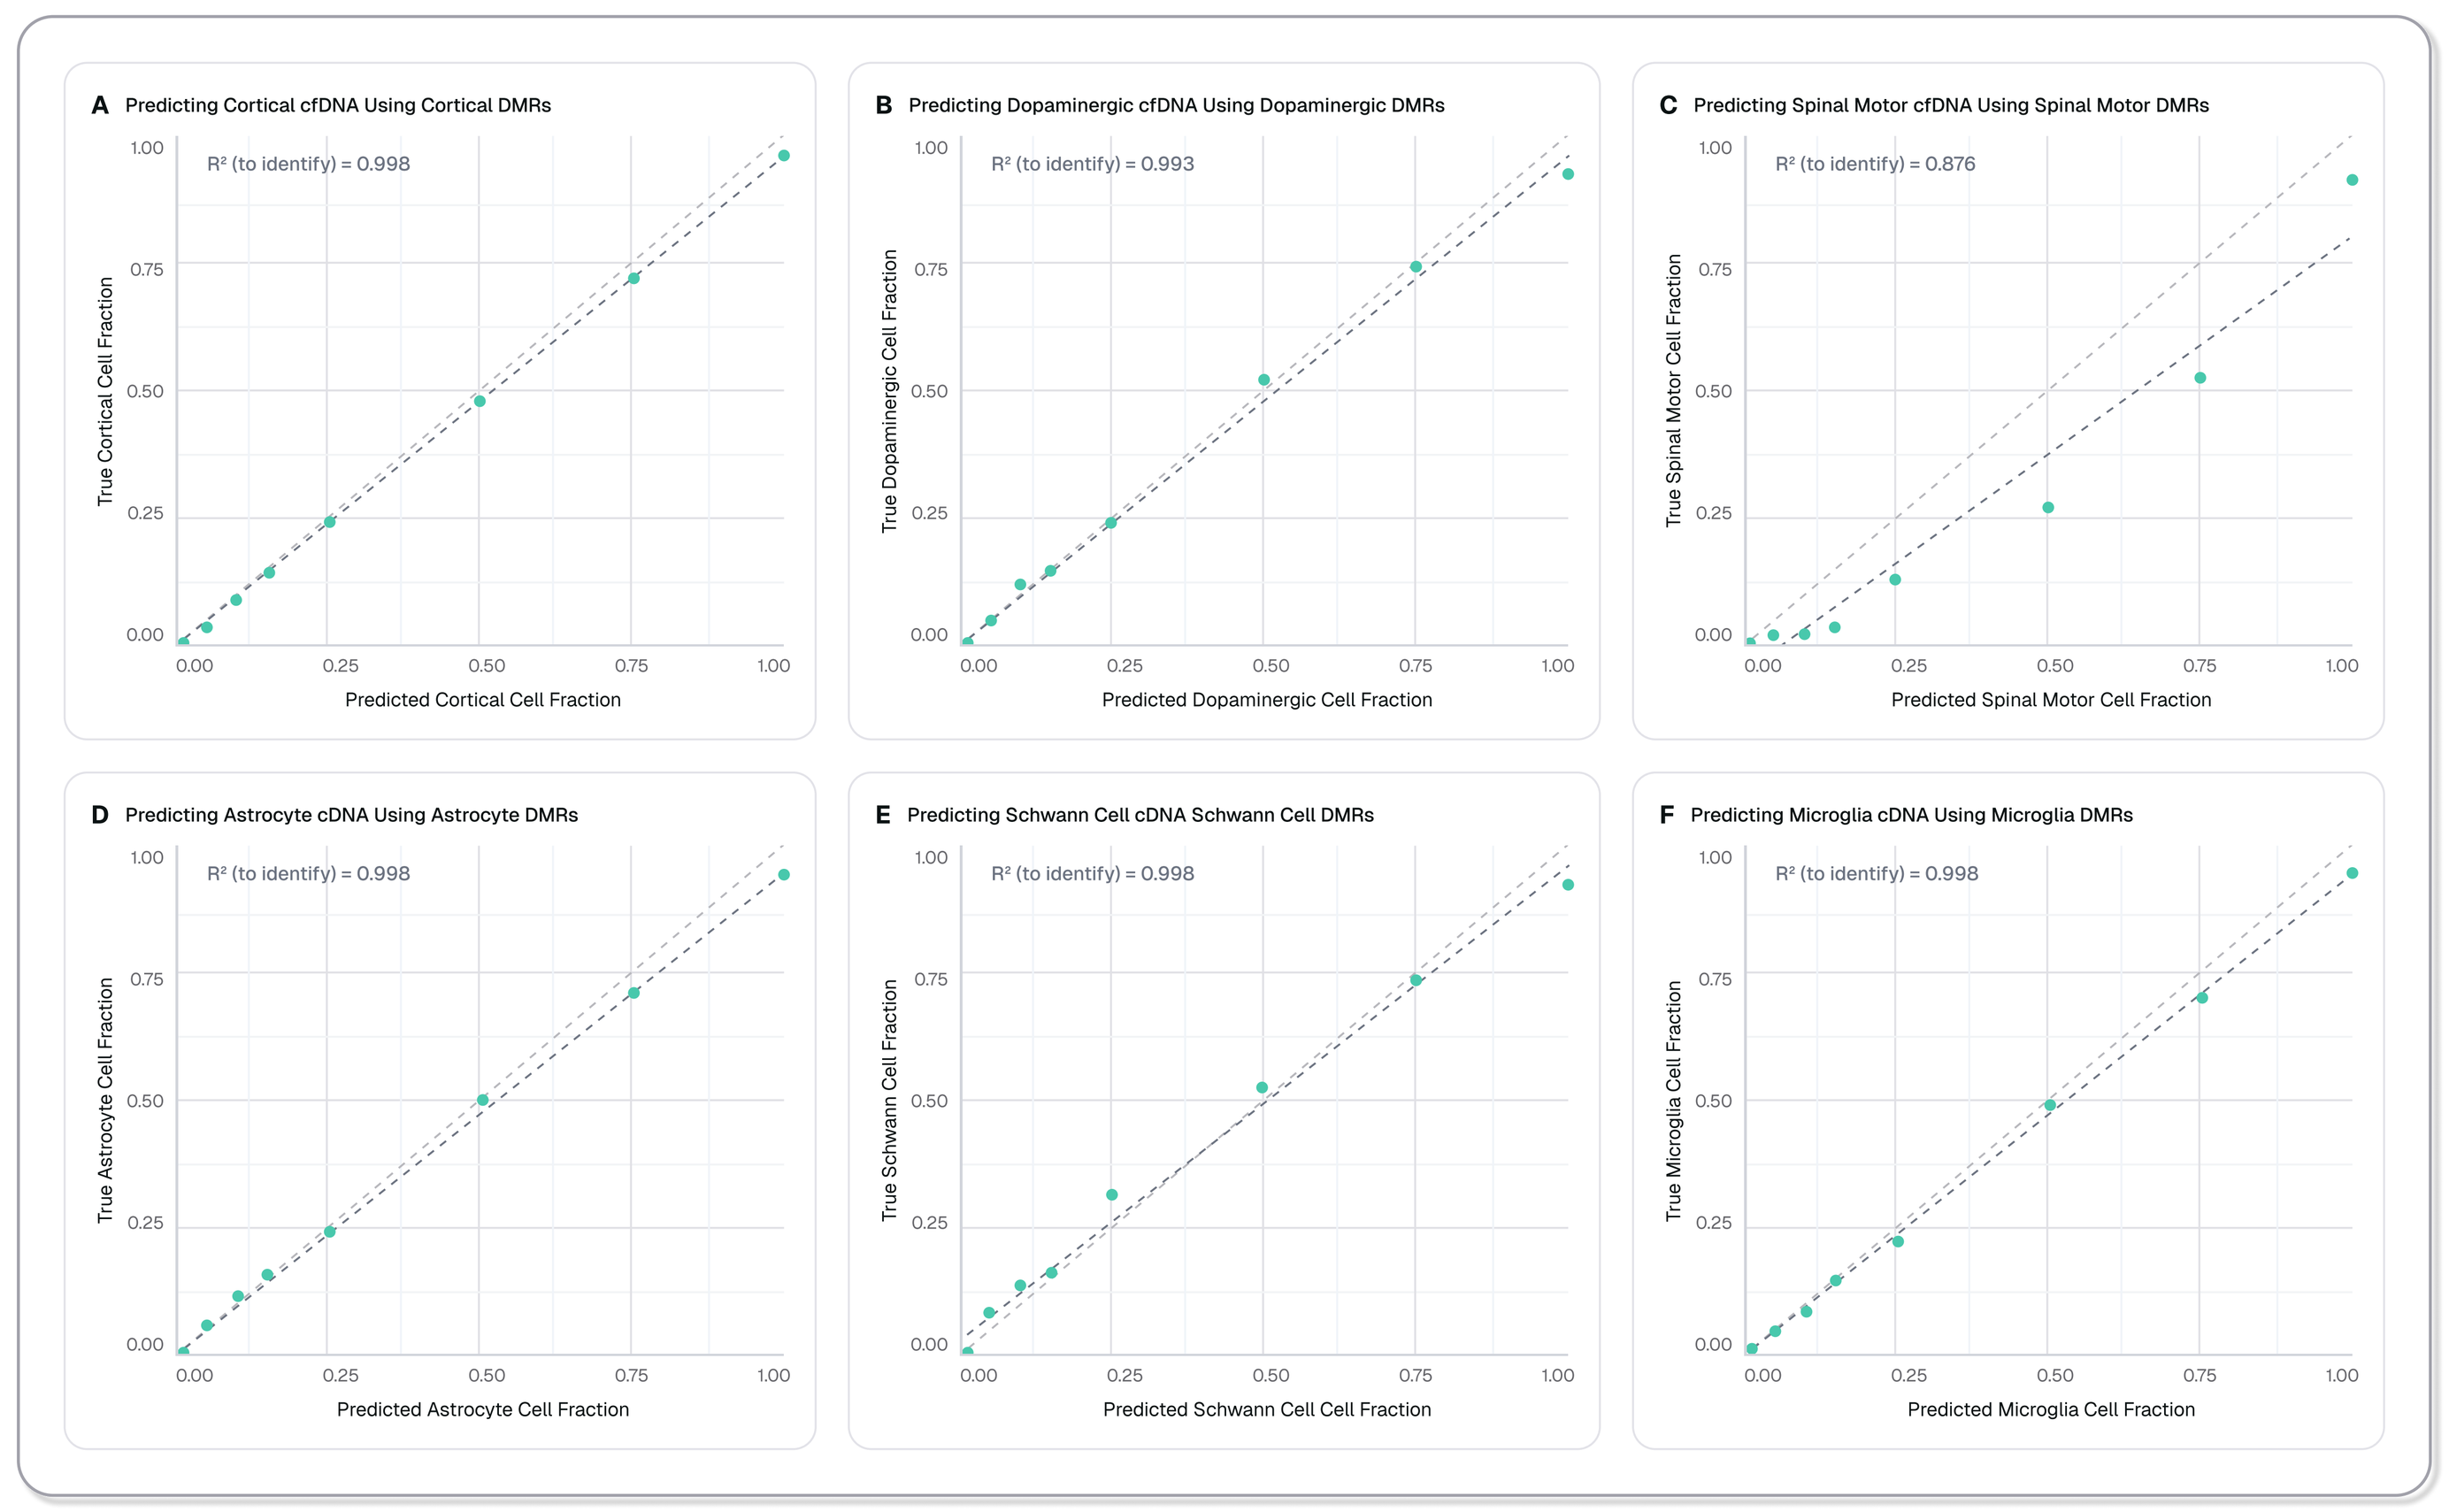
A Predicting Cortical cfDNA Using Cortical DMRs
R² (to identify) = 0.998
B Predicting Dopaminergic cfDNA Using Dopaminergic DMRs
R² (to identify) = 0.993
C Predicting Spinal Motor cfDNA Using Spinal Motor DMRs
R² (to identify) = 0.876
D Predicting Astrocyte cDNA Using Astrocyte DMRs
E Predicting Schwann Cell cDNA Schwann Cell DMRs
F Predicting Microglia cDNA Using Microglia DMRs
R² (to identify) = 0.998

Figure 2. In silico performance demonstrates strong linearity across biologically relevant DNA concentrations. Primary cortical (A, R2 = 0.998), dopaminergic (B, R2 = 0.993), and spinal motor neuron (C, R2 = 0.876) classifiers reliably detected cell-derived DNA in plasma-like backgrounds.

Predicting Disease from Individual Neuron Signals

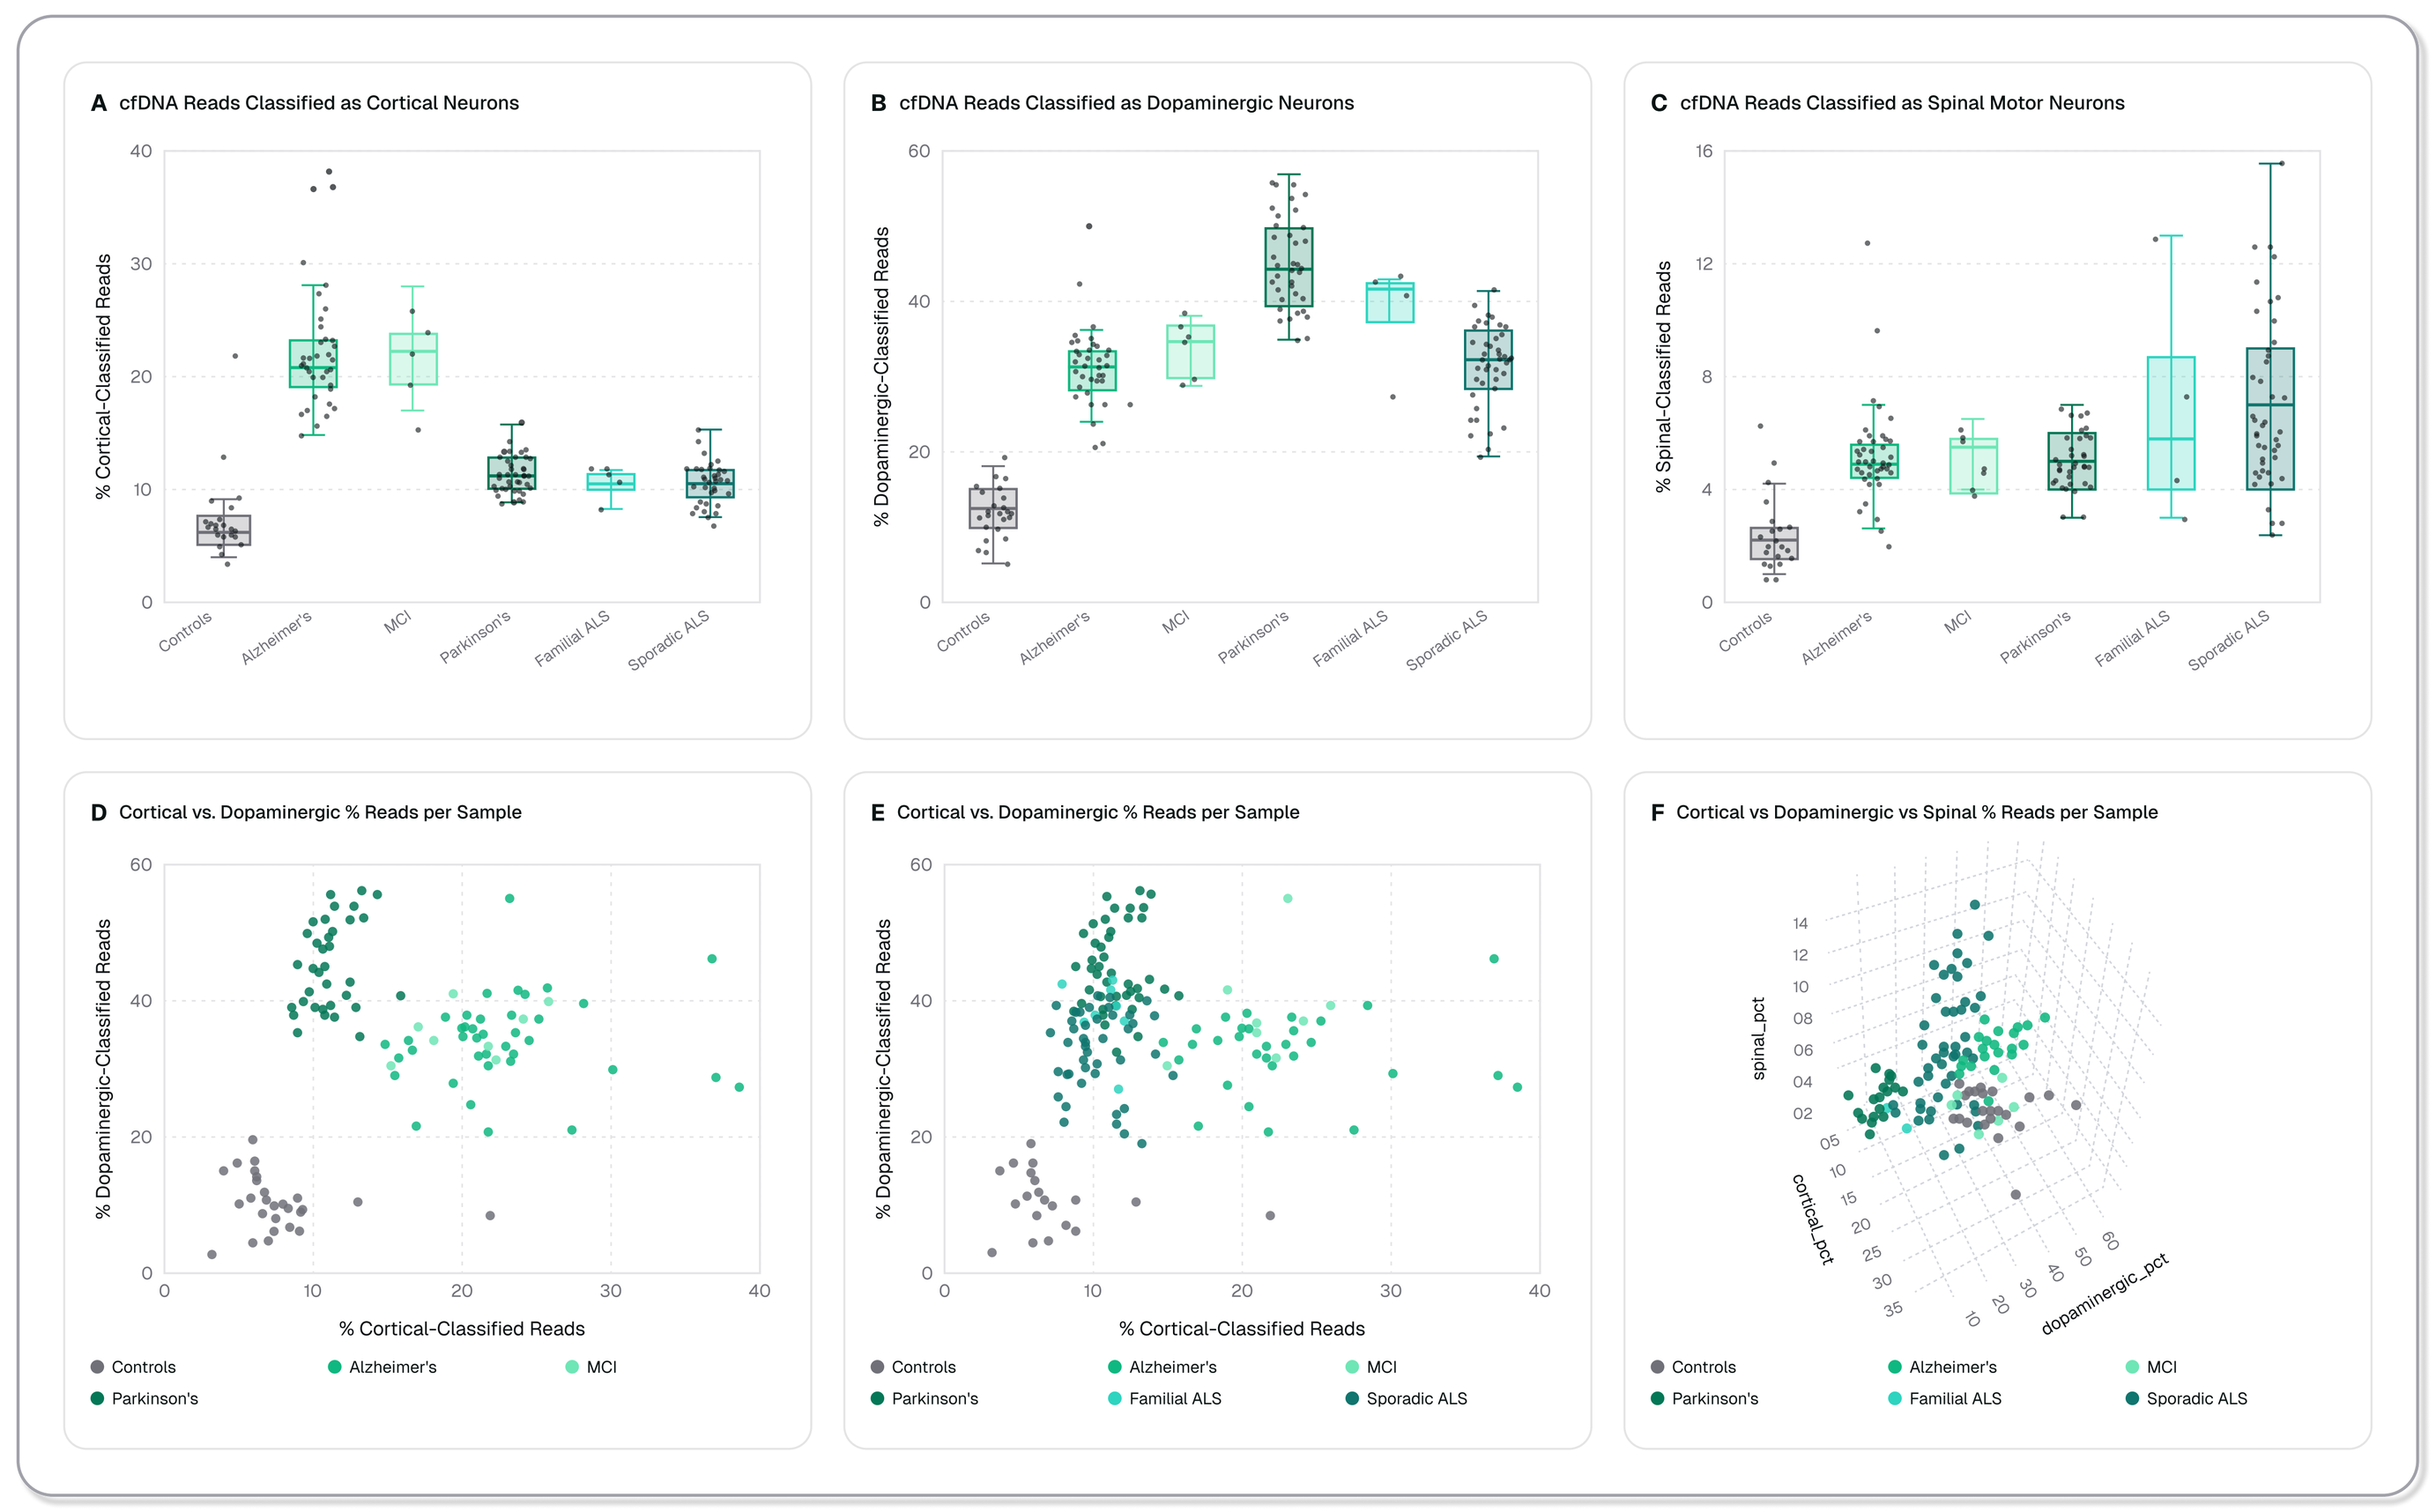
A cfDNA Reads Classified as Cortical Neurons
B cfDNA Reads Classified as Dopaminergic Neurons
C cfDNA Reads Classified as Spinal Motor Neurons
D Cortical vs. Dopaminergic % Reads per Sample
E Cortical vs. Dopaminergic % Reads per Sample
F Cortical vs Dopaminergic vs Spinal % Reads per Sample
Controls
Alzheimer's
MCI
Parkinson's
Familial ALS
Sporadic ALS

Figure 3. Neuron-aligned cfDNA methylation signals distinguish neurodegenerative disease groups. Cortical (A), dopaminergic (B), and spinal motor neuron (C) classifiers showed elevated signals in AD/MCI, PD, and ALS, respectively, consistent with selectively vulnerable neuronal populations.

Predicting Disease from Combined Neuron Signals

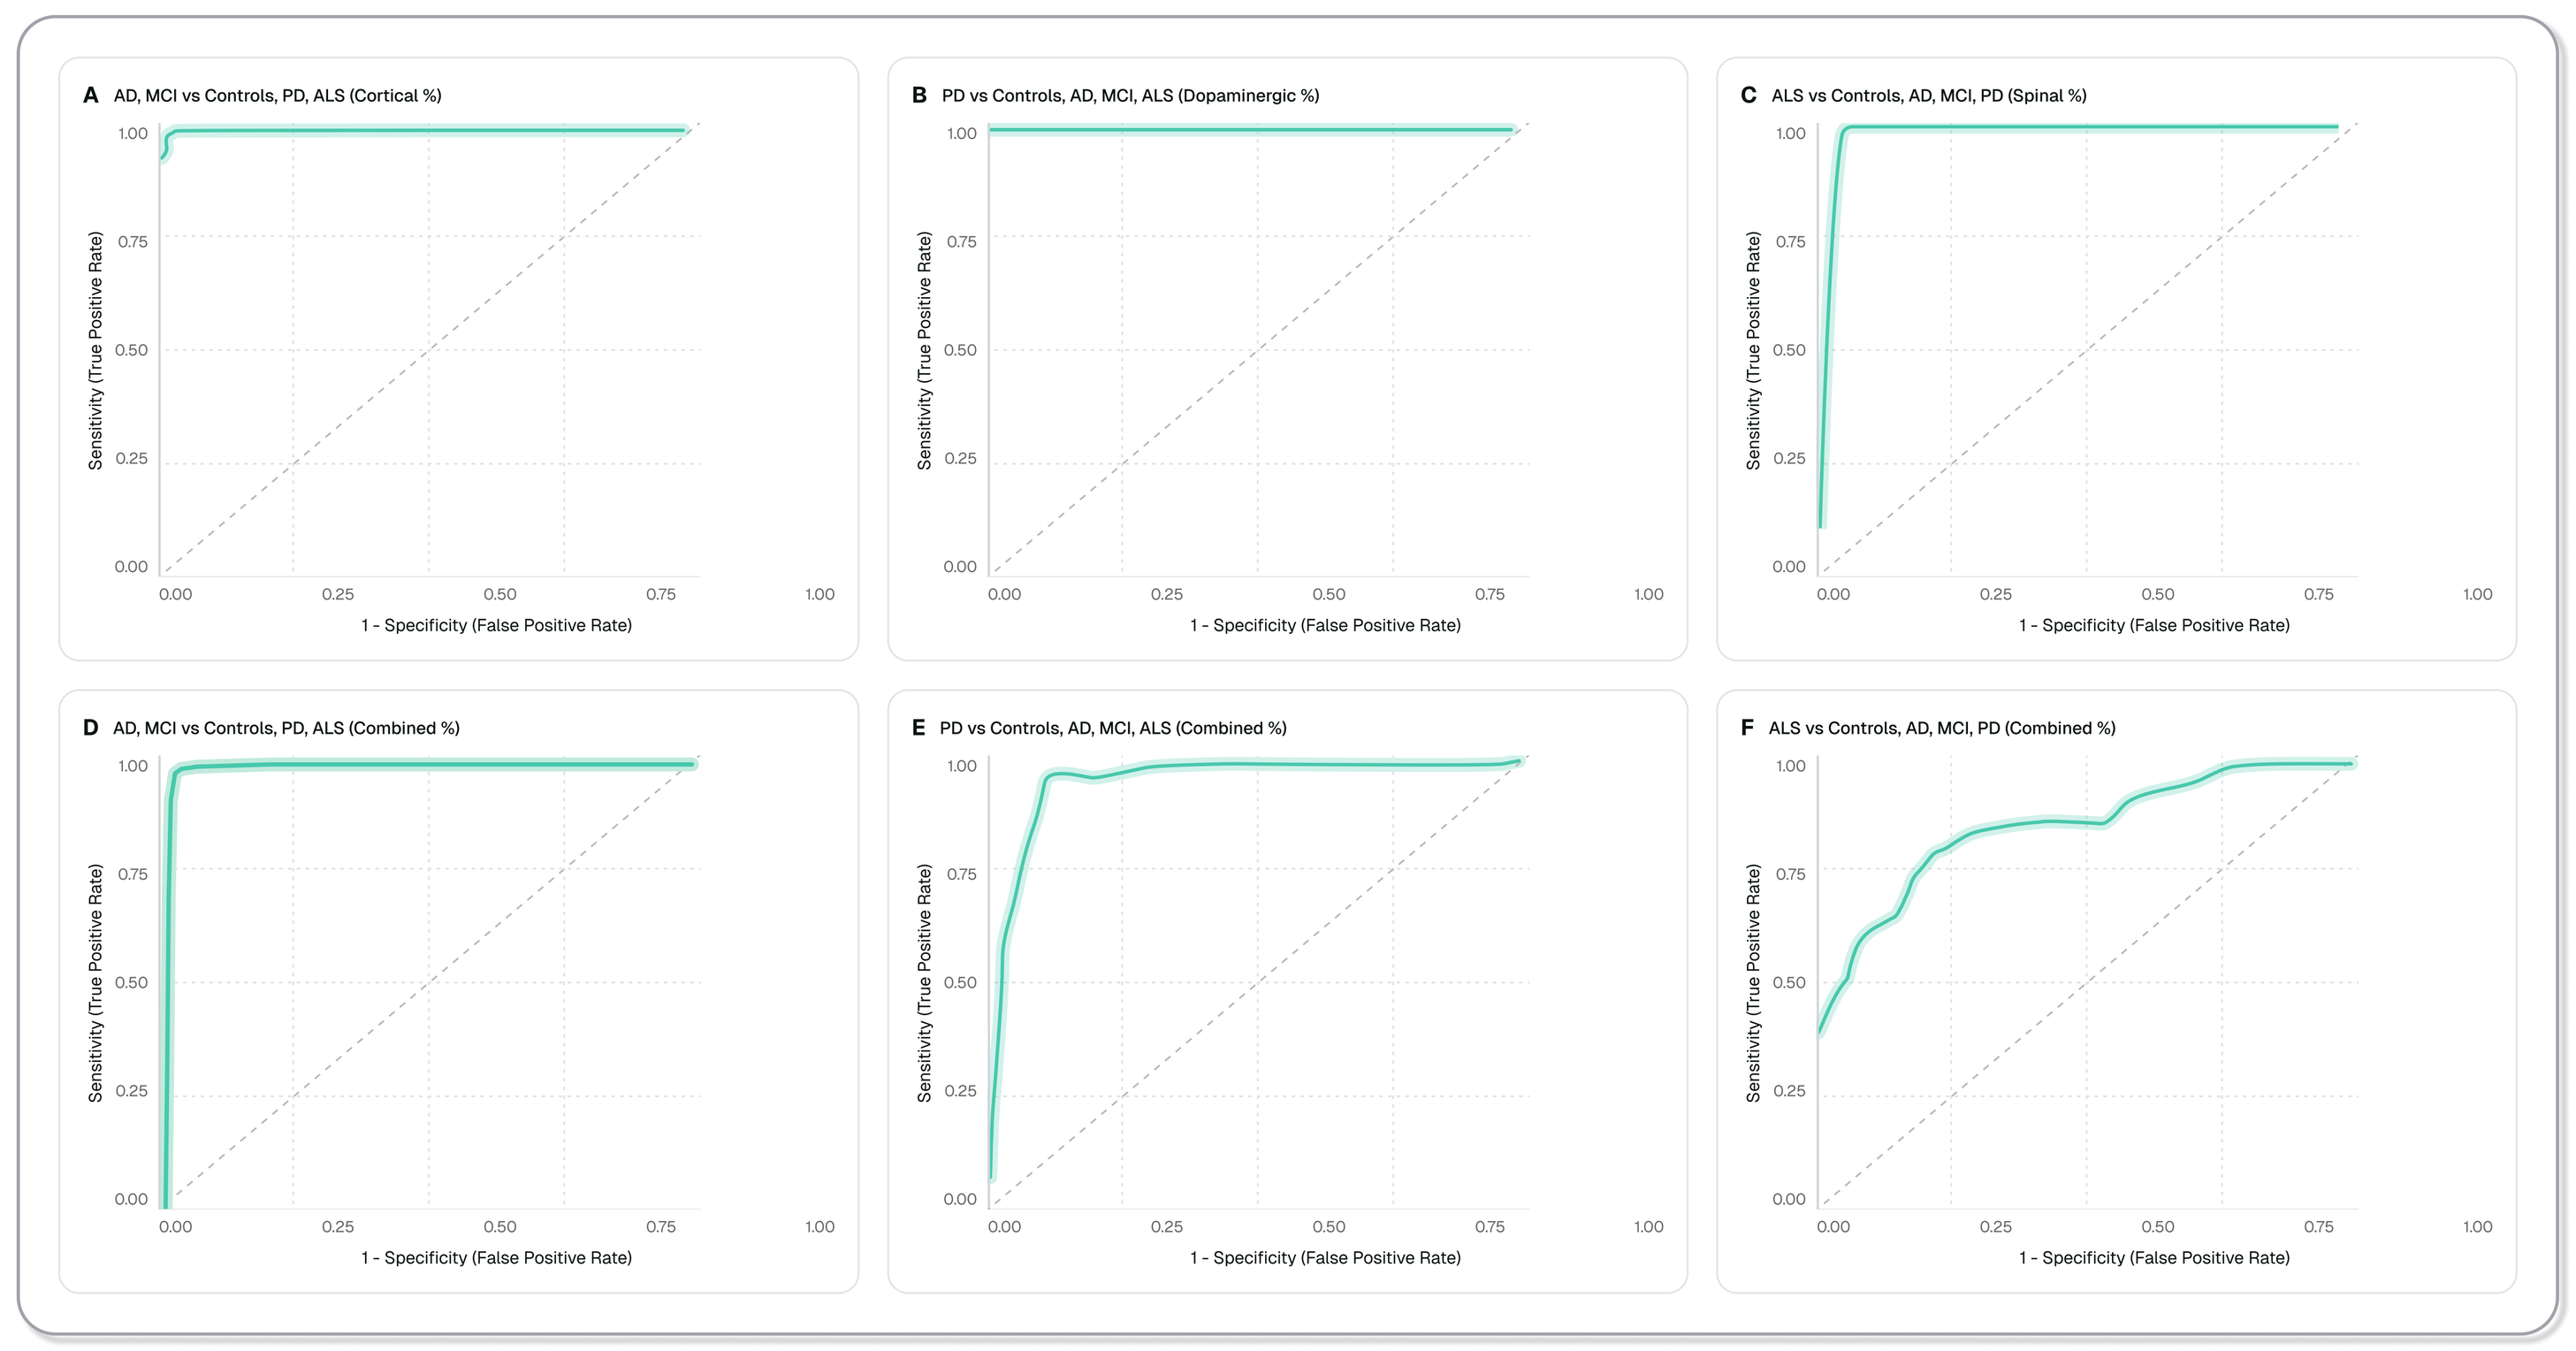
A AD, MCI vs Controls, PD, ALS (Cortical %)
B PD vs Controls, AD, MCI, ALS (Dopaminergic %)
C ALS vs Controls, AD, MCI, PD (Spinal %)
D AD, MCI vs Controls, PD, ALS (Combined %)
E PD vs Controls, AD, MCI, ALS (Combined %)
F ALS vs Controls, AD, MCI, PD (Combined %)
Sensitivity (True Positive Rate)
1 - Specificity (False Positive Rate)

Figure 4. Classifiers combining signatures from multiple neuron types predict disease in multi-disease contexts. Combined neuron classifiers integrating cortical, dopaminergic, and spinal motor neuron-like methylation signatures were evaluated in plasma samples from individuals with AD, PD, ALS, and healthy controls. ROC analyses demonstrated strong disease-versus-control performance (A-C) and maintained discrimination in multi-disease comparisons (D-F), supporting integrated neuron-associated cfDNA methylation profiling for neurodegeneration

Conclusions

Native nanopore methylation sequencing supports proof-of-concept plasma cfDNA deconvolution for neuron-aligned signal detection in neurodegenerative disease. In a filtered plasma cohort of 137 samples, this framework recovered disease-associated cfDNA methylation patterns aligned with selectively vulnerable neuronal populations and supported strong binary and informative multi-disease classification.

Future Studies: Larger longitudinal studies will be required to determine whether cfDNA methylation deconvolution can support presymptomatic detection, disease progression tracking, and therapeutic monitoring in neurodegenerative disease. Continued atlas expansion and benchmarking in independent cohorts will be important next steps. A 775-participant study is currently underway in collaboration with the University of Kansas Alzheimer's Disease Research Center to evaluate these applications in Alzheimer's disease.

References

Pollard C, Aston K, Emery BR, Hill J, Jenkins T. Detection of neuron-derived cfDNA in blood plasma: A new diagnostic approach for neurodegenerative conditions. Frontiers in Neurology. 2023 Oct 31;14:1272960.